Next-Generation, Inducible IL-7–Expressing, Tumor-Infiltrating Lymphocytes by Lentiviral Vector Genetic Modification for Clinical Application

Michael Douglas Crowther,[1] Marie Christine Wulff Westergaard,[1] Phillip Debnam,[2] Victoria Anderson,[2] George R. Pope,[2] Laura Quinn,[2] Emily Schmidt,[2] Adel Toth,[2] Anoop Chandran,[2] Zhaohui Li,[2] Joseph Sanderson,[2] Stine Kiær Larsen,[1] Marco Donia,[1] Özcan Met,[1] Inge Marie Svane[1]

[1]National Center for Cancer Immune Therapy (CCIT-DK), Copenhagen University Hospital, Herlev, Denmark; [2]Adaptimmune, Abingdon, Oxfordshire, UK

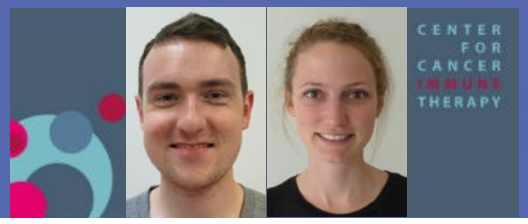
CENTER
FOR
CANCER
IMMUNE
THERAPY

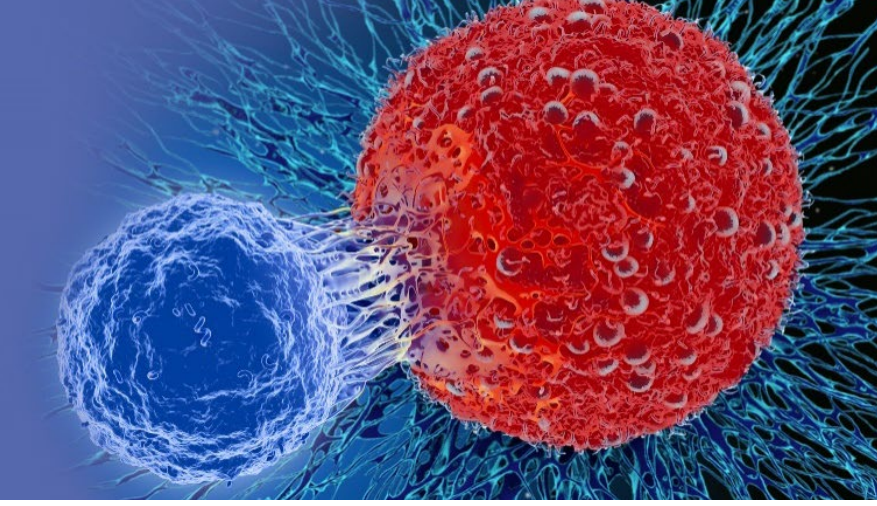

Introduction

- Tumor-infiltrating lymphocyte (TIL) therapy has shown some of the most favorable responses in refractory metastatic melanoma, possibly due to the immunogenicity of this cancer.[1,2]
- TILs are isolated from a patient's own tumor tissues, rapidly expanded in vitro, and then adoptively transferred back into the patient.
- These TILs can recognize and attack cancer cells in large numbers with high specificity.[3]
- In multiple Phase 1/2 trials of melanoma, TIL therapy has demonstrated a robust response rate of up to 50%; however, durable complete responses were only seen in about 15% of treated patients.[4-10]
- Interleukin 7 (IL-7) is a potent stimulator of T-cell proliferation and survival. However, T-cells are incapable of producing their own IL-7 and rely on secretion from surrounding stromal cells.[11]
- We hypothesize that the introduction of an inducible IL-7 gene to TILs (next-generation TIL; ADP-TILIL7; **Figure 1**) will improve their ability to engraft, proliferate, and survive, while maintaining diverse TIL specificity.
- This may translate into improved clinical activity and durability of response when given to patients.

Figure 1. Hypothesized Mechanism of Action of TILs Expressing IL-7: ADP-TILIL7

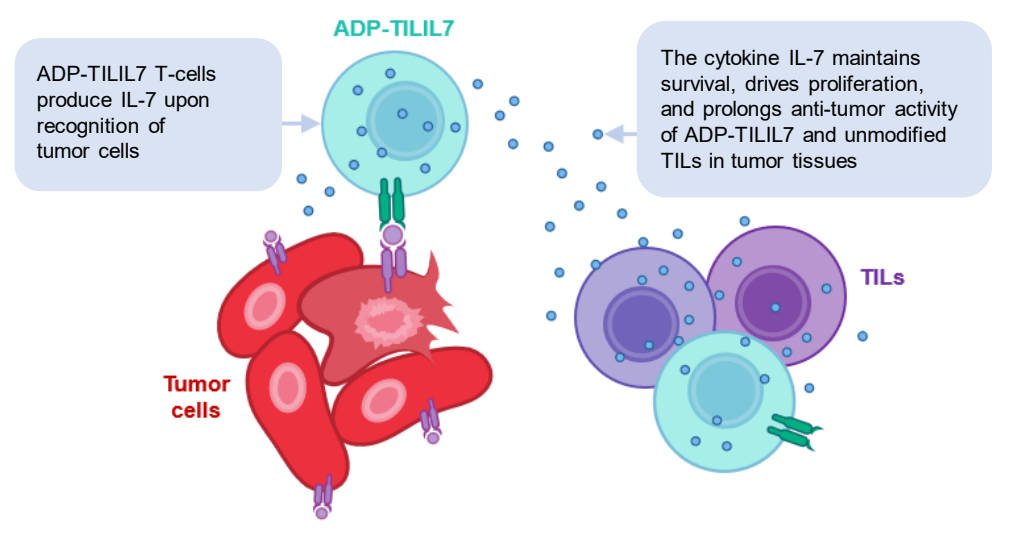
ADP-TILIL7 T-cells produce IL-7 upon recognition of tumor cells
ADP-TILIL7
The cytokine IL-7 maintains survival, drives proliferation, and prolongs anti-tumor activity of ADP-TILIL7 and unmodified TILs in tumor tissues
TILs
Tumor cells

IL-7, interleukin 7; TIL, tumor infiltrating lymphocyte.

- To investigate the ability of IL-7 to enhance T-cell functionality, we first used CD3+ T-cells from healthy donor peripheral blood transduced with an affinity-enhanced T-cell receptor (TCR) specific for the cancer testis antigen melanoma-associated antigen 4 (MAGE-A4) combined with IL-7 under the control of a nuclear factor of activated T-cells (NFAT) inducible promoter as a model system (ADP-A2M4IL7).
 - We found that production of inducible IL-7 by ADP-A2M4IL7 led to improved T-cell expansion (**Figure 2A**) and retention of functionality (**Figure 2B**) upon repeated antigen stimulation in vitro, compared with non-transduced cells and cells transduced with the MAGE-A4-targeted TCR only (ADP-A2M4), without impacting immediate T-cell activity or T-cell specificity.
- Next, we focused on introducing the human IL-7 gene under the control of the same NFAT inducible promoter used in the model system into patient-derived TILs to generate T-cells that secrete IL-7 upon recognition of their cognate antigen.
- Data supporting clinical evaluation of ADP-TILIL7 are presented here.

Figure 2. Production of Inducible IL-7 by ADP-A2M4IL7 Led to Improved T-cell Expansion and Retention of Functionality upon Repeated Antigen Stimulation

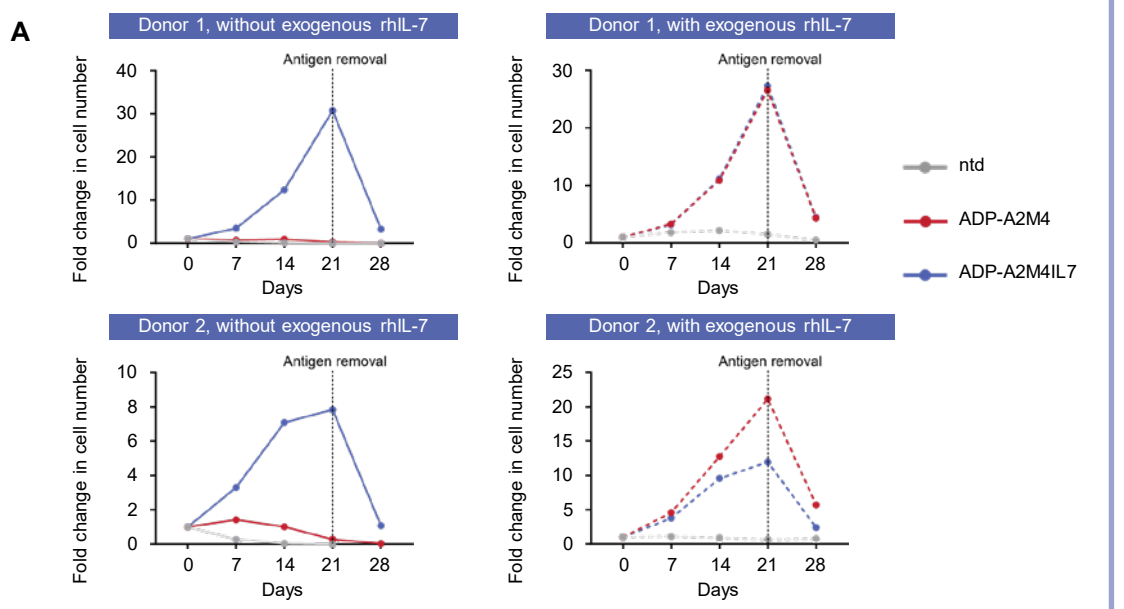
A
Donor 1, without exogenous rhIL-7
Donor 1, with exogenous rhIL-7
Donor 2, without exogenous rhIL-7
Donor 2, with exogenous rhIL-7
Antigen removal
Fold change in cell number
Days
ntd
ADP-A2M4
ADP-A2M4IL7

- ADP-A2M4, ADP-A2M4IL7, and ntd T-cells from 2 donors were stimulated weekly with irradiated A375 (MAGE-A4 positive) cells in the absence (solid lines) or presence (dotted lines) of 20 ng/mL exogenous rhIL-7.
- A375 (MAGE-A4 positive) cells were removed on Day 21.
- Cell counts were performed every 7 days, and total cell number was plotted as fold change relative to total cell number at the start of the assay.

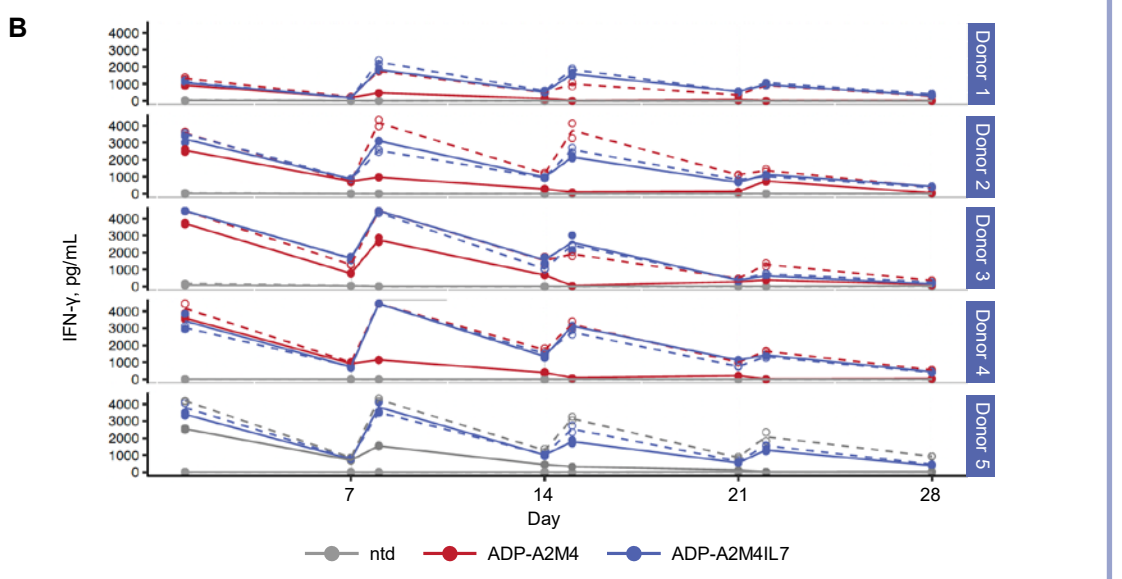
B
IFN-γ, pg/mL
Donor 1
Donor 2
Donor 3
Donor 4
Donor 5
Day
ntd
ADP-A2M4
ADP-A2M4IL7

- ADP-A2M4, ADP-A2M4IL7, and ntd T-cells were stimulated weekly with irradiated A375 (MAGE-A4 positive) cells in the absence (solid lines) or presence (dotted lines) of 20 ng/mL exogenous rhIL-7.
- Supernatants were collected at Day 1, 7, 8, 14, 21, 22, and 28.
- IFNγ levels were determined by Luminex and MAGPIX (Luminex, Austin, TX).
- Data are faceted by donor; icons represent the values at each time point for each donor.

IFNγ, interferon gamma; IL-7, interleukin 7; ntd, non-transduced; rhIL-7, recombinant human interleukin 7.

Development of ADP-TILIL7

- Young TILs from patient donors were isolated and expanded, followed by activation, IL-7 transduction, and implementation of a rapid expansion protocol (REP; **Figure 3**).
- We tested the functionality of the lentiviral vector by determining the production of IL-7 upon non-specific stimulation and found that ADP-TILIL7 reliably secreted IL-7.
 - Transducing the TILs during culture showed a 68% reduction in IL-7 secretion and 92% reduction in vector copy number compared to transducing the TILs before the REP.

Figure 3. Overview of IL-7 Transduction in Young TILs

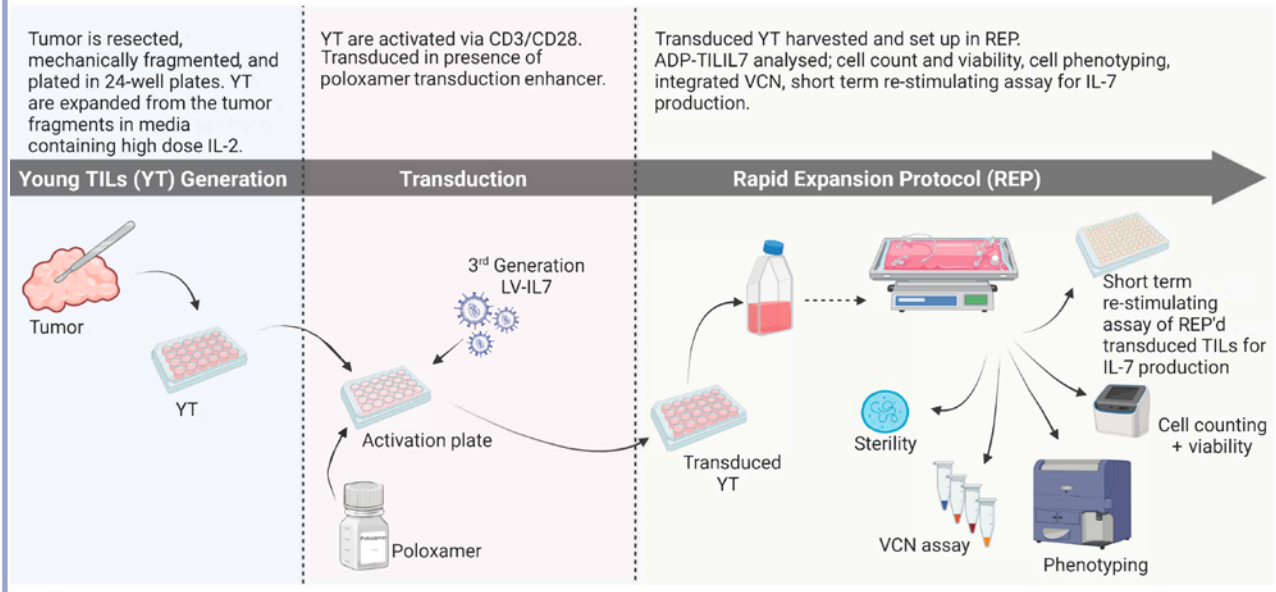
Tumor is resected, mechanically fragmented, and plated in 24-well plates. YT are expanded from the tumor fragments in media containing high dose IL-2.
YT are activated via CD3/CD28. Transduced in presence of poloxamer transduction enhancer.
Transduced YT harvested and set up in REP. ADP-TILIL7 analysed; cell count and viability, cell phenotyping, integrated VCN, short term re-stimulating assay for IL-7 production.
Young TILs (YT) Generation
Transduction
Rapid Expansion Protocol (REP)
Tumor
YT
3rd Generation LV-IL7
Activation plate
Poloxamer
Transduced YT
Sterility
VCN assay
Phenotyping
Short term re-stimulating assay of REP'd transduced TILs for IL-7 production
Cell counting + viability

IL-7, interleukin 7; LV, lentiviral vector; MOI, multiplicity of infection; REP, rapid expansion protocol; TIL, tumor-infiltrating lymphocyte; VCN, vector copy number; YT, young TILs.

ADP-TILIL7 Functional Evaluation

- In short-term restimulation assays measuring IL-7 secretion, we found:
 - ADP-TILIL7 T-cells reliably secreted IL-7 at a similar concentration to that of ADP-A2M4IL7 T-cells (**Figure 4A**), which had been shown to increase survival.
 - IL-7 production increased when transduction occurred in the presence of a poloxamer transduction enhancer (**Figure 4B**).

Figure 4. ADP-TILIL7 Reliably Secrete IL-7 in Response to TCR Stimulation

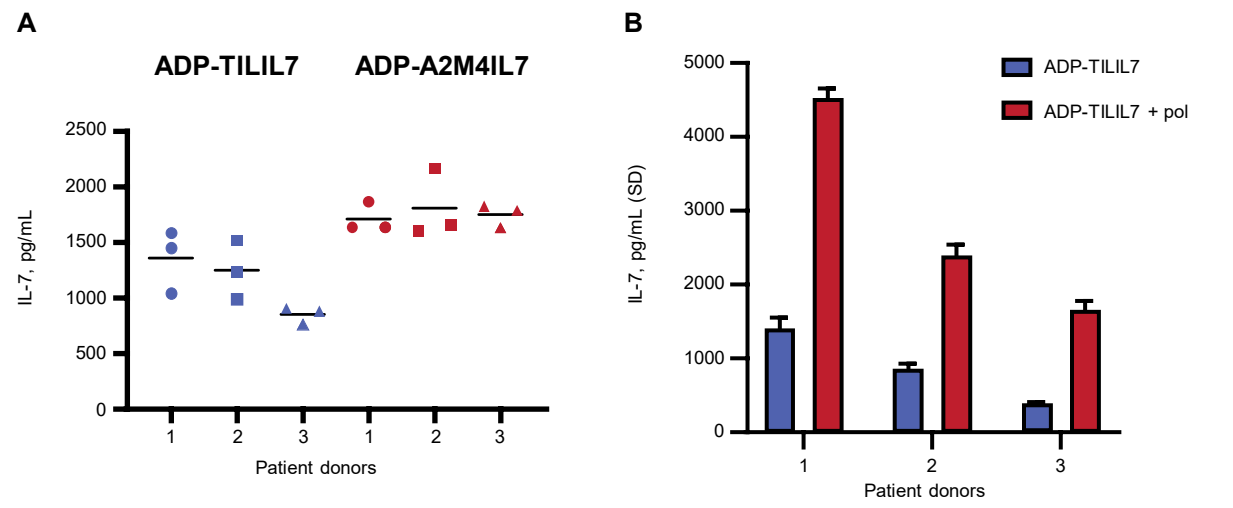
A
ADP-TILIL7
ADP-A2M4IL7
IL-7, pg/mL
Patient donors
B
ADP-TILIL7
ADP-TILIL7 + pol
IL-7, pg/mL (SD)
Patient donors

- (A) YT were transduced with the IL-7 lentiviral vector and subsequently rapidly expanded. Transduced REP TILs were then activated in a short-term restimulation assay alongside PBMC-derived CD3+ cells transduced with a MAGE-A4-targeting TCR plus IL-7 gene lentiviral vector. (B) YT transduced with IL-7 lentiviral vector in the presence or absence of a pol, and subsequently rapidly expanded. Transduced REP TILs were then activated in a short-term restimulation assay.
- IL-7 secretion was measured via ELISA.

ELISA, enzyme-linked immunosorbent assay; IL-7, interleukin 7; MAGE-A4, melanoma-associated antigen A4; PBMC, peripheral blood mononuclear cell; pol, poloxamer transduction enhancer; REP, rapid expansion protocol; TCR, T-cell receptor; TIL, tumor-infiltrating lymphocyte; YT, young TILs.

- Transduction did not negatively impact expansion or phenotype as evidenced by comparisons between non-transduced and transduced TILs on Day 14 of REP:
 - Similar fold expansion (**Figure 5A**).
 - Similar percentages of central memory, effector memory, naive, and TEMRA T-cells out of total CD3+ T-cells, based on expression of CCR7 and CD45RA (**Figure 5B**).
 - Similar expression of IL-7 receptor α (CD127; **Figure 5C**).

Figure 5. Transduction of Young TILs Did Not Negatively Impact Expansion or Phenotype

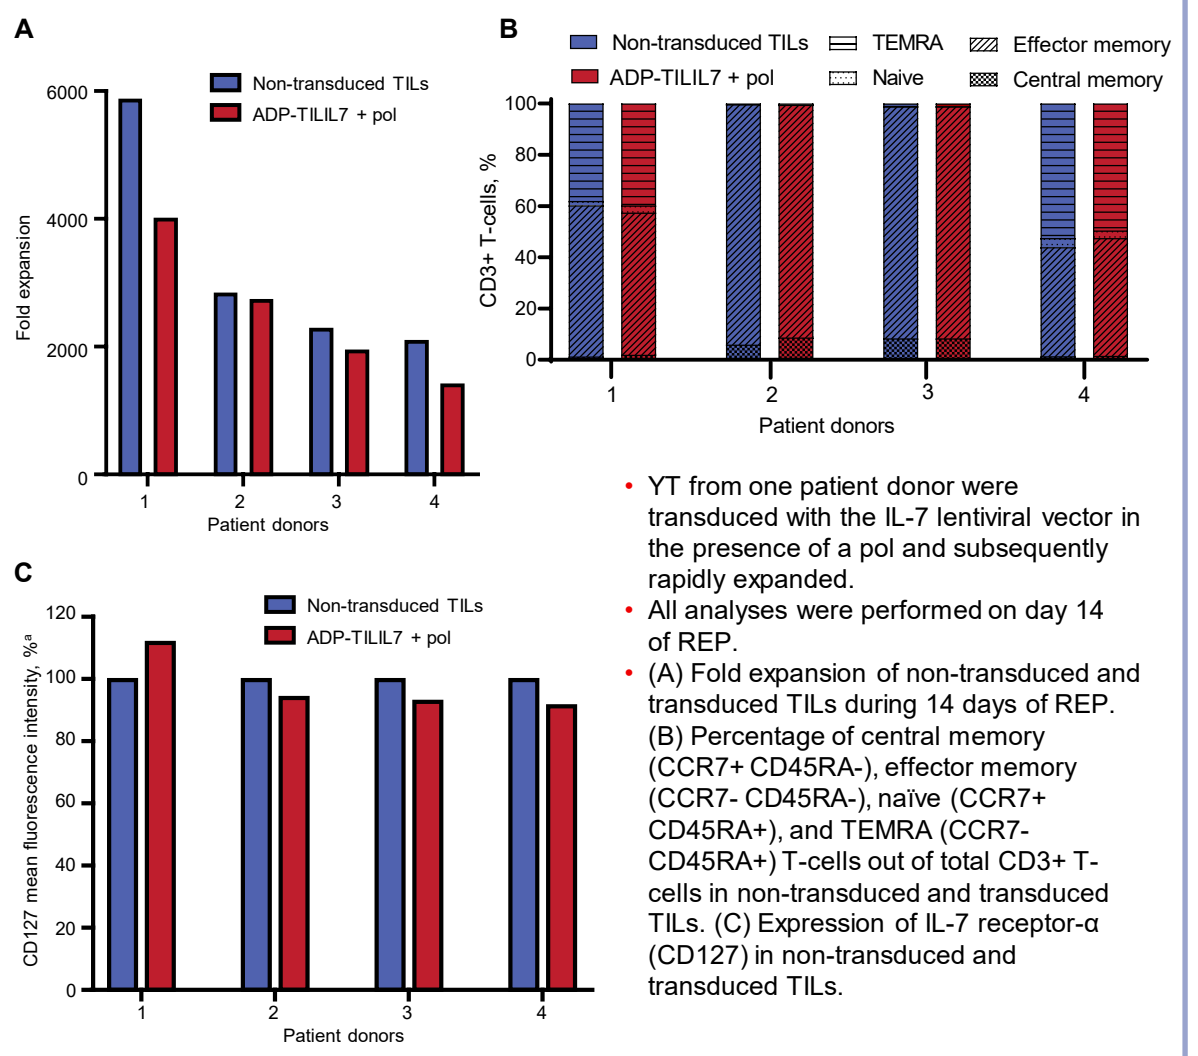
A
Fold expansion
Non-transduced TILs
ADP-TILIL7 + pol
Patient donors
B
Non-transduced TILs
ADP-TILIL7 + pol
TEMRA
Naive
Effector memory
Central memory
CD3+ T-cells, %
Patient donors
C
CD127 mean fluorescence intensity, %a
Non-transduced TILs
ADP-TILIL7 + pol
Patient donors

- YT from one patient donor were transduced with the IL-7 lentiviral vector in the presence of a pol and subsequently rapidly expanded.
- All analyses were performed on day 14 of REP.
- (A) Fold expansion of non-transduced and transduced TILs during 14 days of REP. (B) Percentage of central memory (CCR7+ CD45RA-), effector memory (CCR7- CD45RA-), naïve (CCR7+ CD45RA+), and TEMRA (CCR7- CD45RA+) T-cells out of total CD3+ T-cells in non-transduced and transduced TILs. (C) Expression of IL-7 receptor-α (CD127) in non-transduced and transduced TILs.

[a]Mean fluorescence intensity values were normalized to the expression on non-transduced TILs. TCR, T-cell receptor; IL-7, interleukin 7; pol, poloxamer transduction enhancer; REP, rapid expansion protocol; TIL, tumor-infiltrating lymphocyte; YT, young TILs.

Conclusions

- We have developed a reliable transduction method for young TILs utilizing lentiviral vector in the presence of a poloxamer transduction enhancer.
- The ADP-TILIL7 final product produces biologically relevant amounts of IL-7, which has been shown in vitro to maintain survival and prolong proliferation of T-cells.
- Based on our preclinical studies, a single-center, Phase 1 clinical trial will be initiated with ADP-TILIL7 to treat patients with metastatic melanoma.

M-216
American Society of Gene + Cell Therapy (ASGCT), May 16–19, 2022; Hybrid, Washington, DC, USA

References

1. Hulen TM, et al. Immuno. 2021;1(3):194. 2. Muul LM, et al. J Immunol. 1987;138(3):989. 3. June CH, et al. Sci Transl Med. 2015;7(280):280. 4. Rosenberg SA, et al. Clin Cancer Res. 2011;17(13):4550. 5. Andersen R, et al. Ann Oncol. 2018;29(7):1575. 6. Ellebaek E, et al. J Transl Med. 2012;10:169. 7. Forget M-A, et al. Clin Cancer Res. 2018;24:4416. 8. Dafni U, et al. Ann Oncol. 2019;30(12):1902. 9. Besser MJ, et al. Clin Cancer Res. 2013;19(17):4792. 10. Dudley ME, et al. J Immunother. 2002;25(3):243. 11. Mackall CL, et al. Nat Rev Immunol. 2011;11(5):330.

Abbreviations Used in Text

IL-7, interleukin 7; MAGE-A4, melanoma-associated antigen A4; NFAT, nuclear factor of activated T-cells; REP, rapid expansion protocol; TCR, T-cell receptor; TIL, tumor-infiltrating lymphocyte.

Acknowledgements and Disclosures

Writing assistance was provided by Gabrielle Knafler, MSc, PhD, of Excel Scientific Solutions, which was contracted and compensated by Adaptimmune for these services. Figure 3 was created with BioRender.com. Michael Douglas Crowther (michael.douglas.crowther@regionh.dk): Research funding and travel expenses from Adaptimmune.