Updated Data from Cohorts 1 and 2 of an Ongoing Study with ADP-A2AFP SPEAR T-Cells

Tim Meyer[1], Bruno Sangro[2], Amit Mahipal[3], Lipika Goyal[4], Matthew J. Frigault[5], Debashis Sarker[6], Lynn G. Feun[7], Petr F. Hausner[8], Mitesh J. Borad[9], Kit Wong[10], Anthony B. El-Khoueiry[11], Richard Finn[12], Ahmed Kaseb[13], Benjamin R. Tan[14], Fiona Thistlethwaite[15], Bassel El-Rayes[16], Jordi Bruix[17], Joana Senra[18], Thejo Annareddy[18], Trupti Trivedi[18], Svetlana Fayngerts[18], Jenny Sampson[18], Elliot Norry[18]

[1]University College, London, UK; [2]Clinica Universidad de Navarro, IDISNA and CIBEREHD, Pamplona, Spain; [3]Mayo Clinic, Rochester, MN; [4]Massachusetts General Hospital Cancer Center, Boston, MA; [5]Massachusetts General Hospital Cancer Center, Boston, MA; [6]King's College London, London, UK; [7]Sylvester Comprehensive Cancer Center, Miami, FL; [8]University of Maryland, Baltimore, MD; [9]Mayo Clinic, Phoenix, AZ; [10]Seattle Cancer Alliance, Seattle, WA; [11]University of Southern California, Los Angeles, CA; [12]UCLA Jonsson Comprehensive Cancer Center, Los Angeles, CA; [13]University of Texas MD Anderson Cancer Center, Houston, TX; [14]Washington University School of Medicine, St Louis, MO; [15]Christie NHS Foundation and University of Manchester, Manchester, UK; [16]Emory University School of Medicine, Atlanta, GA; [17]University Hospital of Barcelona, BCLC group, Hospital Clinic, Barcelona, Spain; [18]Adaptimmune, Oxfordshire UK, and Philadelphia, PA, USA

An electronic copy of the poster can be viewed by scanning the QR code here https://bit.ly/3hMvoOI

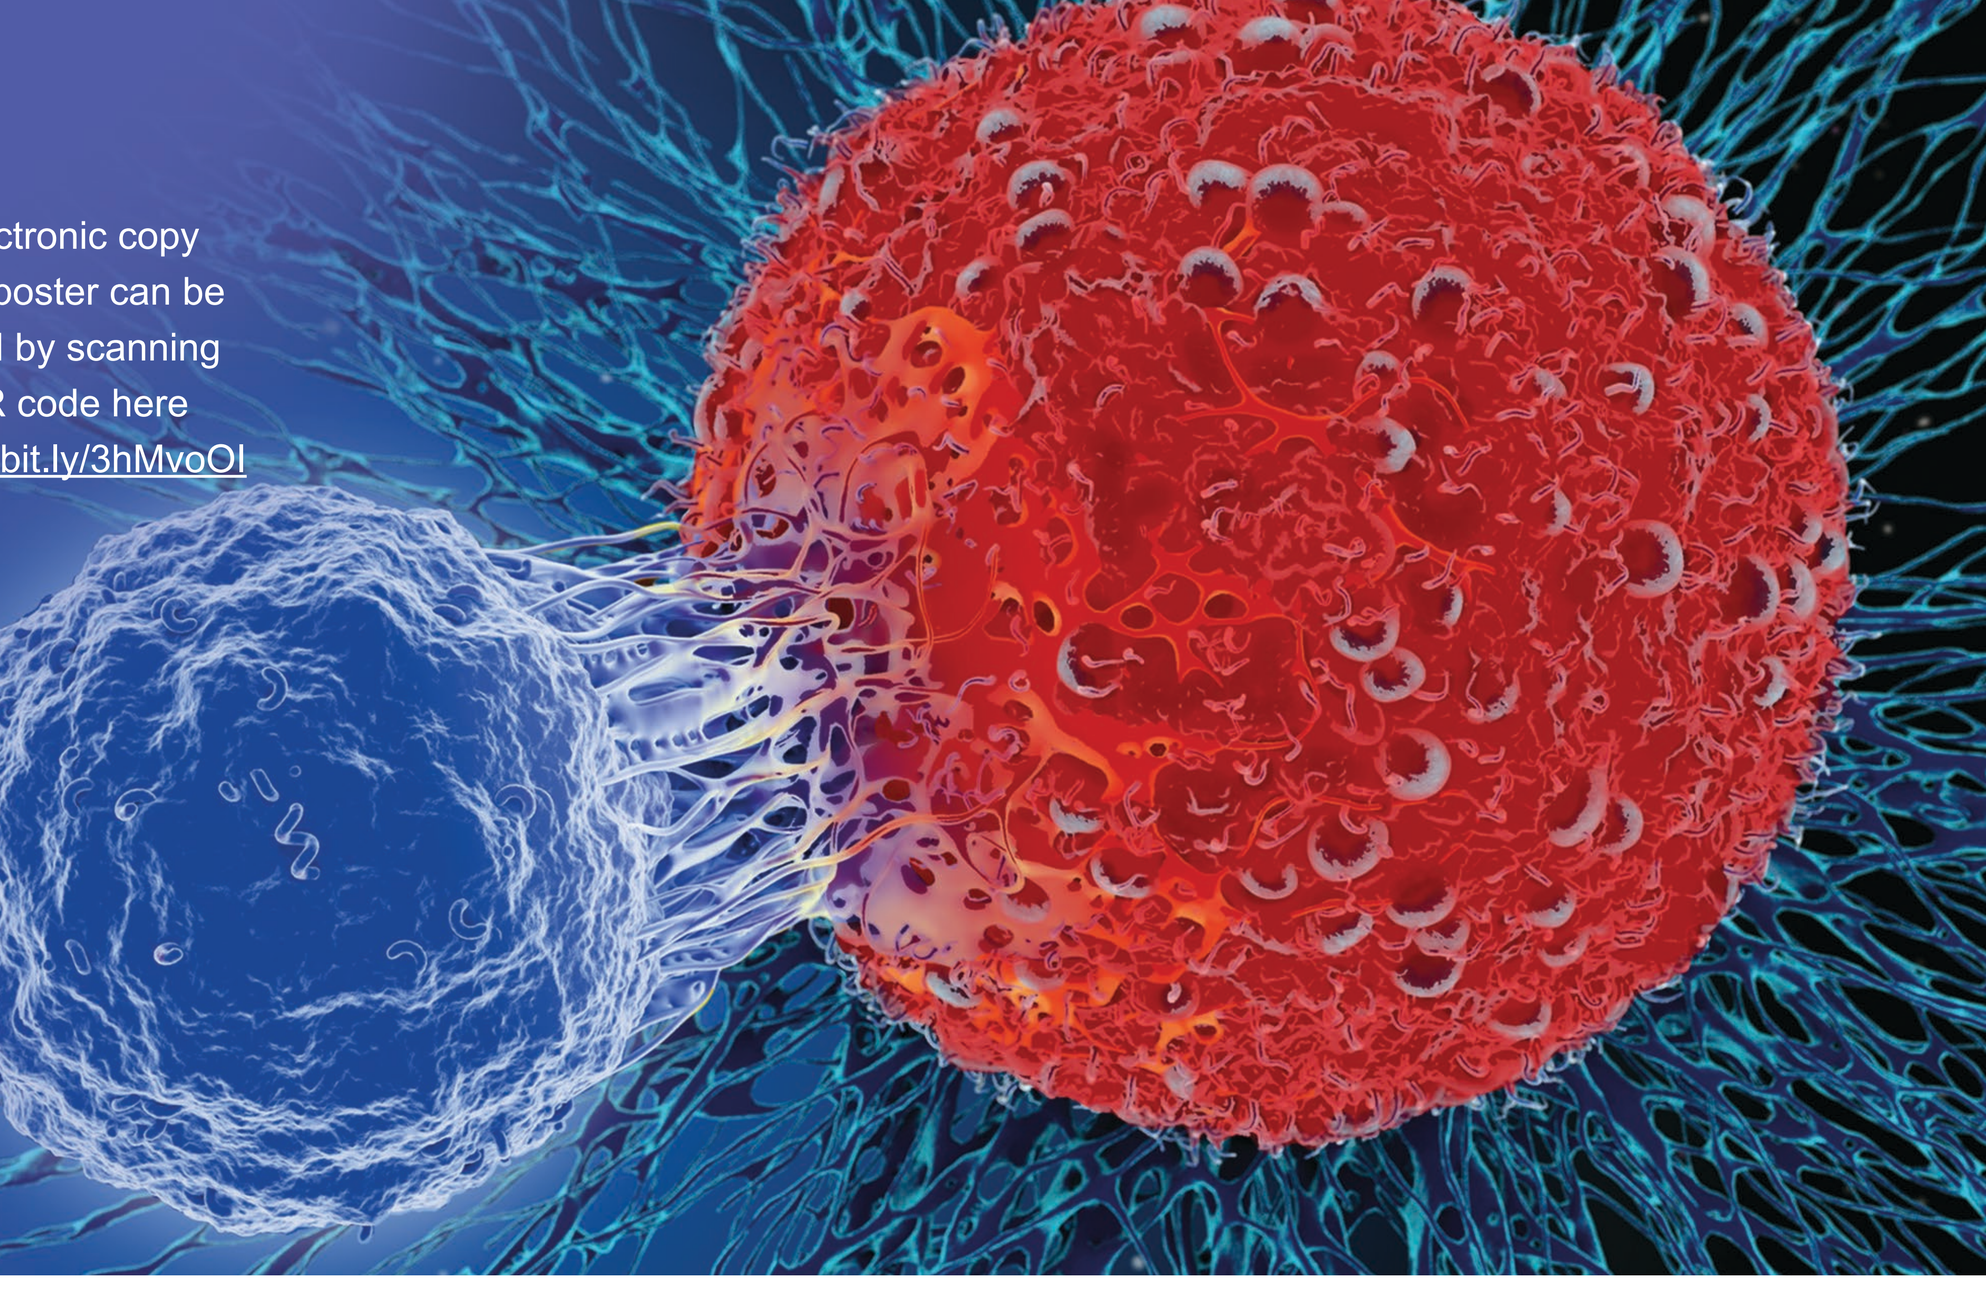

Introduction

- AFP is an abundant serum protein in the fetus that is transcriptionally repressed shortly after birth
- Reappearance of AFP in the circulation of adults is associated with liver regeneration, hepatitis, chronic liver diseases, or malignant growth[1]
- Elevation of serum AFP is associated with poor prognosis in HCC[2]
- HCC is the third highest cause of cancer-related death globally[3]
- There remains considerable unmet need for new therapies for advanced HCC

Objectives

- Evaluate safety and anti-tumor activity in patients with HCC who are being treated with genetically engineered affinity-enhanced autologous SPEAR T-cells (ADP-A2AFP) directed toward the HLA-A*02-restricted AFP peptide FMNKFIYEI4 in an ongoing phase 1 trial (NCT03132792)
- This presentation will address initial safety and anti-tumor activity data from patients in Cohorts 1 and 2 in this ongoing trial[4]

Methods

- This is a first-in-human study in HCC patients who were not amenable to transplant, resection, or loco-regional therapy and failed/intolerant/refused standard-of-care treatment
 - Of note, the protocol has been amended to include patients with non-HCC tumors expressing AFP, in a separate cohort, Cohort 4
- Patients must be HLA-A*02:01⁺ or HLA-A*02:642⁺
- Evidence of tumor or serum AFP expression is required
 - Original protocol required AFP expression by IHC at ≥2+ staining in ≥40% of HCC tumor cells
 - Protocol amended to allow enrollment by either serum AFP ≥400 ng/mL or IHC of ≥1+ in ≥20% of tumor cells
 - Later amended to allow serum AFP ≥100 ng/mL
- Non-cancerous liver tissue must have ≤5% of cells staining for AFP by IHC
- Additional key inclusion criteria are Child-Pugh score ≤6, ECOG ≤1, and adequate organ function
- Up to 35 patients with HCC will be enrolled using a modified 3+3 design

Table 1. Dose escalation

Cohort	No. of Patients	Transduced Cell Doses	Lymphodepletion Regimen	Interval for Safety Review
1	2–6	1 × 10^8 (range: 0.8–1.2 × 10^8)	Cy: 500 mg/m²/d × 3 days Flu: 20 mg/m²/d × 3 days (Low-dose regimen)	21-day stagger between patients
2	3–6	1 × 10^9 (range: 0.5–1.2 × 10^9)	Cy: 500 mg/m²/d × 3 days Flu: 20 mg/m²/d × 3 days (Low-dose regimen)	7-day stagger
3	3–6	5 × 10^9 (range: 1.2–6 × 10^9)	Cy: 600 mg/m²/d × 3 days Flu: 30 mg/m²/d × 4 days (High-dose regimen)	7-day stagger
	HCC Expansion phase: Up to 30 patients total	5 × 10^9 (range: 1.2–10 × 10^9)	Cy: 600 mg/m²/d × 3 days Flu: 30 mg/m²/d × 4 days (High-dose regimen)	No stagger
4	Up to 10 patients with AFP-expressing tumor types other than HCC	5 × 10^9 (range: 1.2–10 × 10^9)	Cy: 600 mg/m²/d × 3 days Flu: 30 mg/m²/d × 4 days (High-dose regimen)	

The trial is currently dosing patients in HCC Expansion and Cohort 4
DLTs and dose escalation as determined by the SRC

Trial Design

Figure 1. Trial design

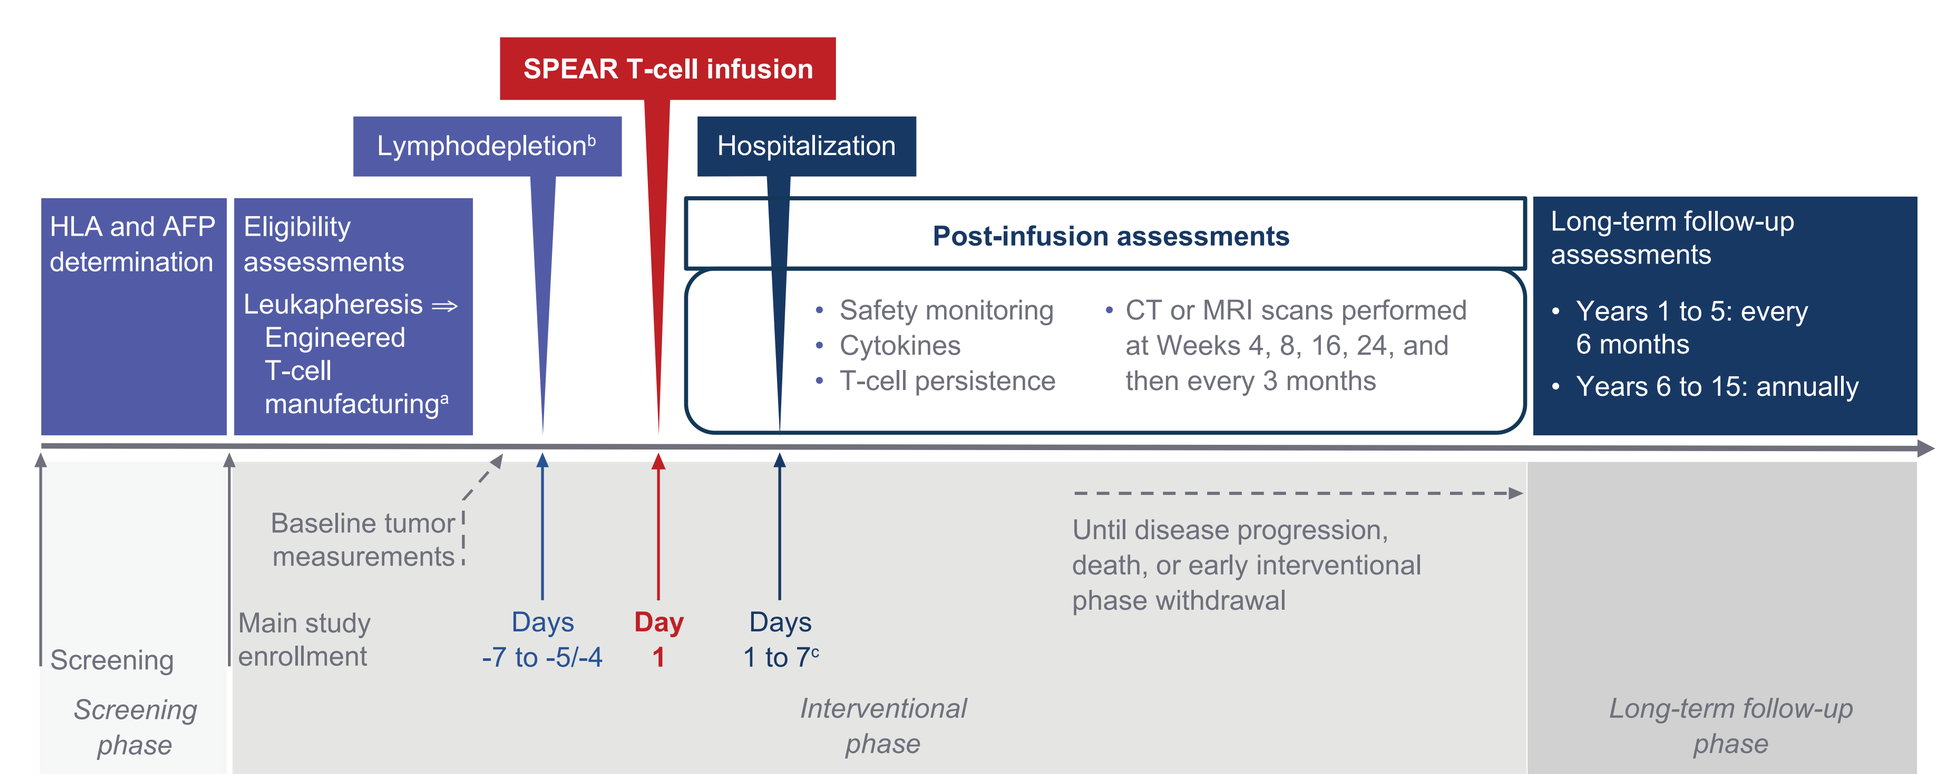
SPEAR T-cell infusion
Lymphodepletion[b]
Hospitalization
HLA and AFP determination
Eligibility assessments
Leukapheresis ⇒ Engineered T-cell manufacturing[a]
Post-infusion assessments
• Safety monitoring
• Cytokines
• T-cell persistence
• CT or MRI scans performed at Weeks 4, 8, 16, 24, and then every 3 months
Long-term follow-up assessments
• Years 1 to 5: every 6 months
• Years 6 to 15: annually
Baseline tumor measurements
Screening
Main study enrollment
Days -7 to -5/-4
Day 1
Days 1 to 7[c]
Until disease progression, death, or early interventional phase withdrawal
Screening phase
Interventional phase
Long-term follow-up phase

[a]T-cell selection; lentiviral gene transfer of affinity-enhanced TCR; T-cell expansion
[b]See Table 1
[c]14 days in the UK

Results

Figure 2. Persistence of transduced T-cells over time

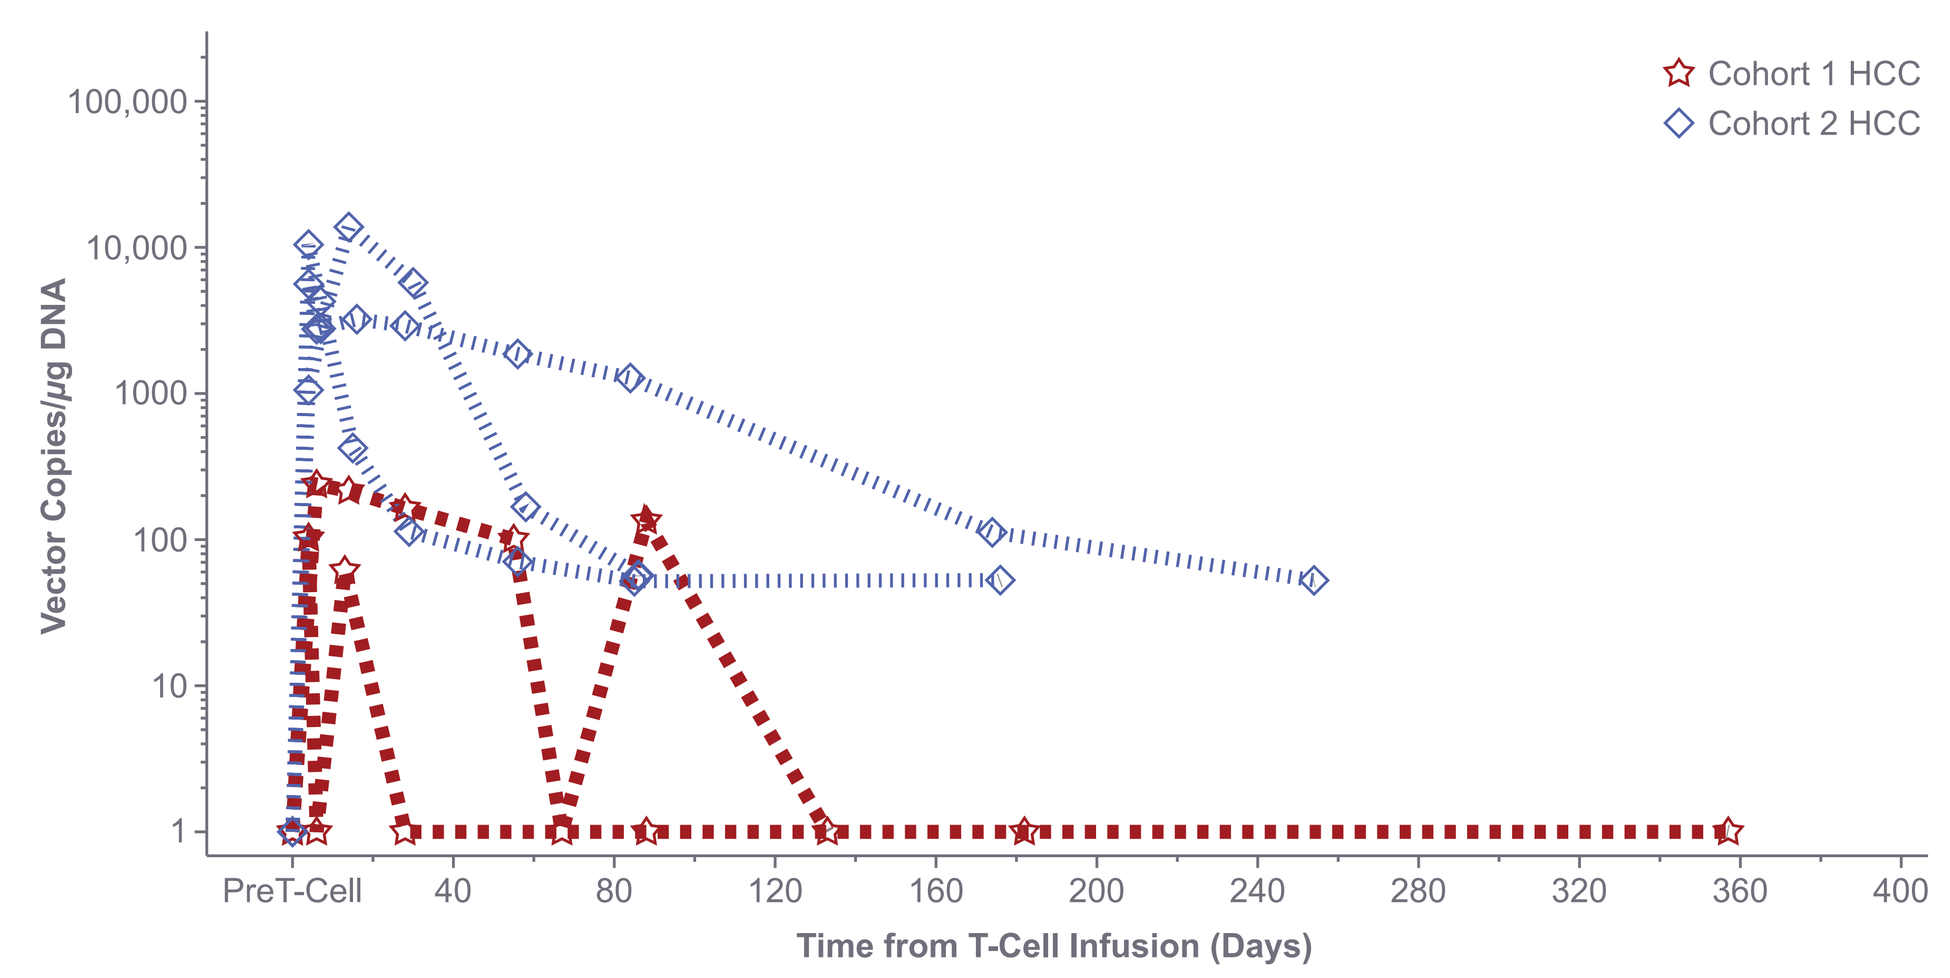
Cohort 1 HCC
Cohort 2 HCC
Vector Copies/μg DNA
100,000
10,000
1000
100
10
1
PreT-Cell
40
80
120
160
200
240
280
320
360
400
Time from T-Cell Infusion (Days)

mITT: all eligible patients who received at least 1 T-cell infusion
y-axis is in log scales
Time from T-cell infusion is calculated as (date of sample collection − date of T-cell) + 1 if sample collection date is equal or greater than date of T-cell; otherwise, time from T-cell infusion is calculated as (date of sample collection − date of T-cell)

- ADP-A2AFP SPEAR T-cells persist in the periphery for several months after infusion
- Peak expansion and SPEAR T-cell persistence appears to be dose dependent
- Persistence of ADP-A2AFP T-cells was measured by qPCR of the PSI element sequence in genomic DNA extracted from PBMCs

Figure 3. Serum AFP expression

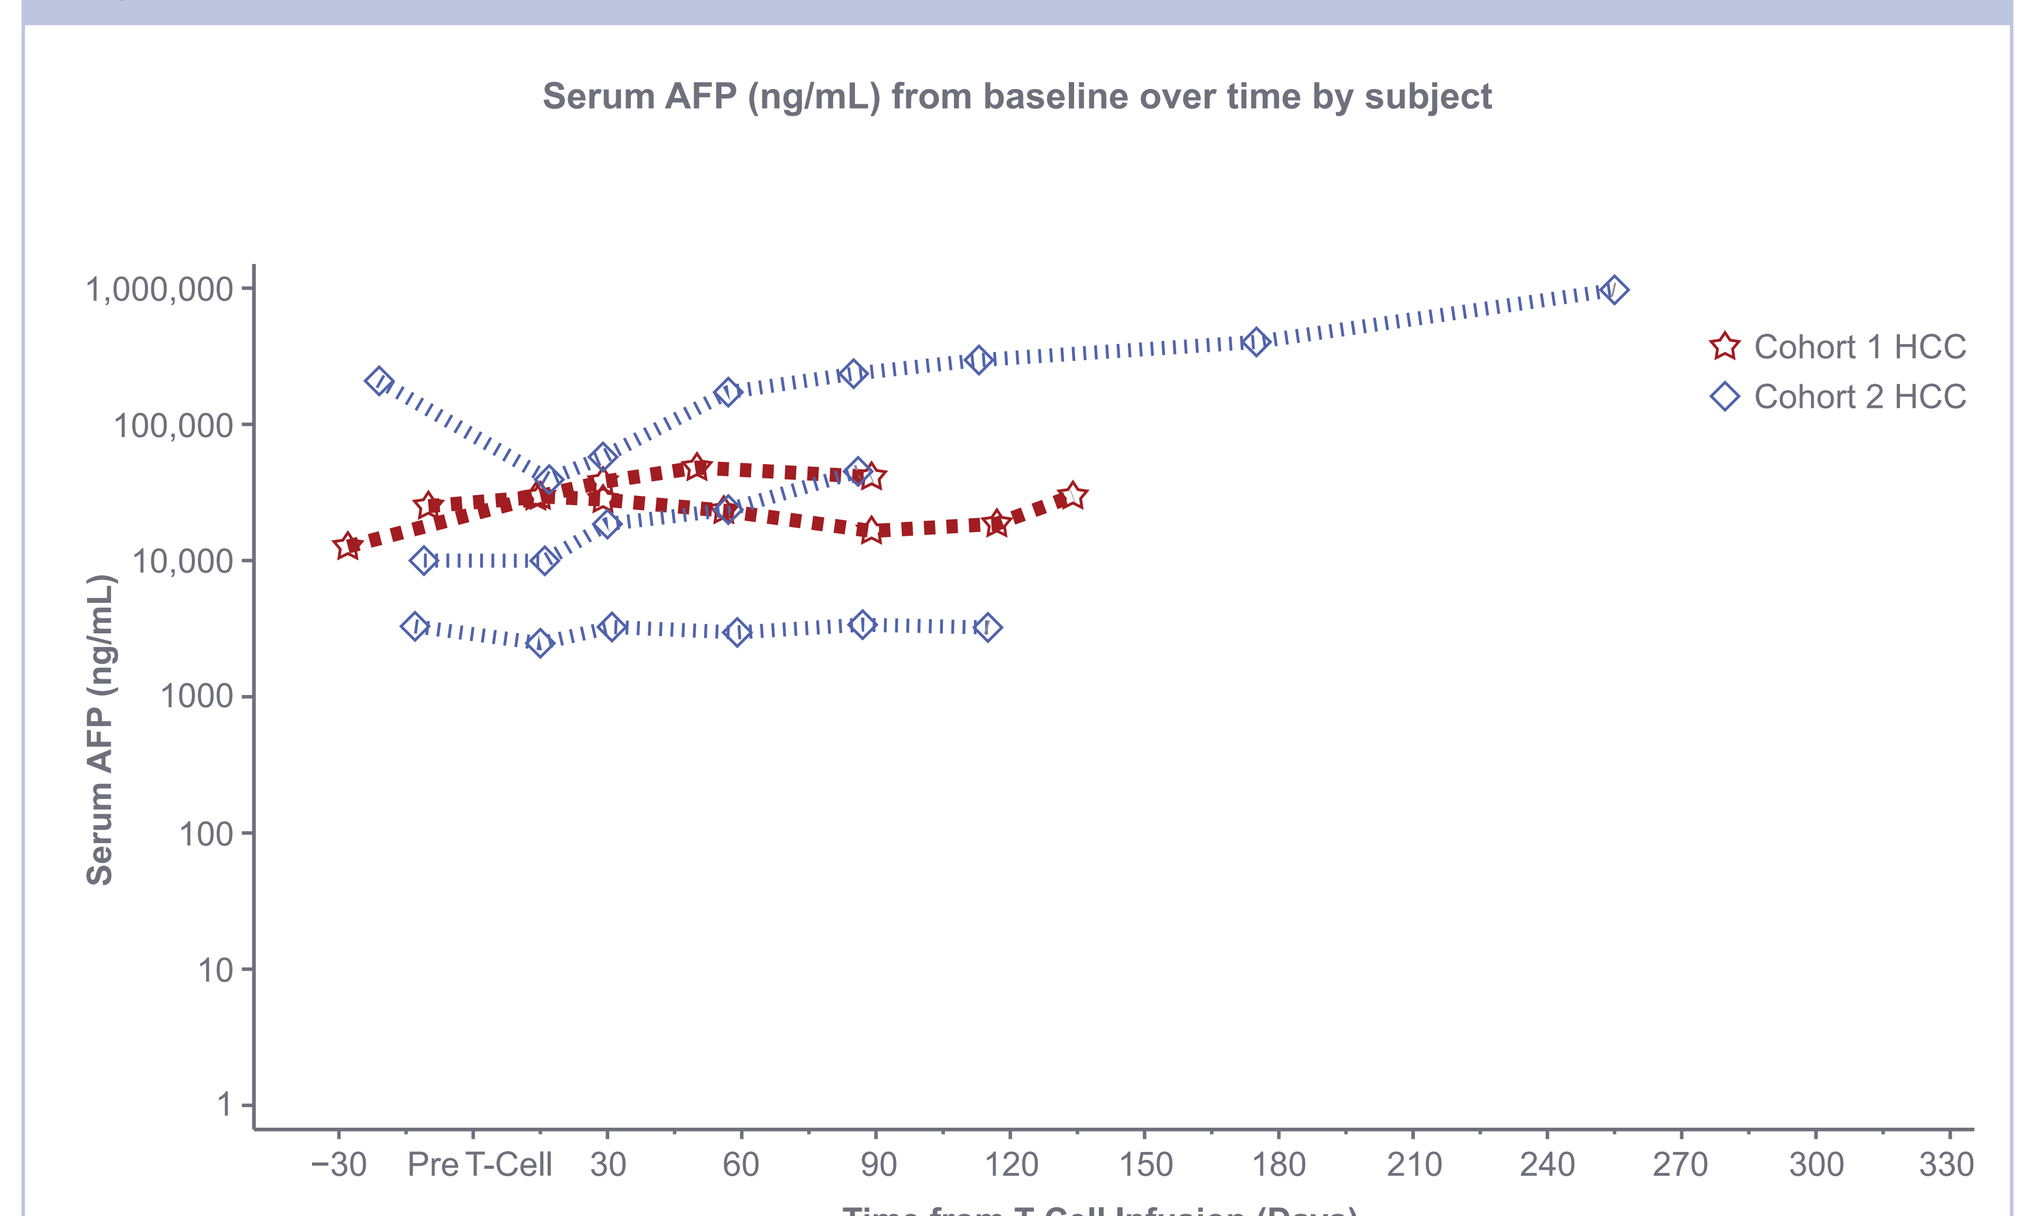
Serum AFP (ng/mL) from baseline over time by subject
Cohort 1 HCC
Cohort 2 HCC
Serum AFP (ng/mL)
1,000,000
100,000
10,000
1000
100
10
1
−30
Pre T-Cell
30
60
90
120
150
180
210
240
270
300
330
Time from T-Cell Infusion (Days)

- Transient decrease in serum AFP was observed in some patients

Table 2. Patient characteristics

Cohort	Patient	Age/Sex	Underlying Liver Disease	Previous Treatments	SPEAR T-Cell Dose
1	1	61/M	HCV cirrhosis treated with antiviral agent and RNA negative	Gemcitabine/cisplatin, sorafenib, tislelizumab	0.1 billion transduced cells
	2	17/M	None	Doxorubicin/cisplatin, gemcitabine/oxaliplatin, pembrolizumab, AutoSynVax, avastin, thalidomide/temodar, XRT lung metastases	0.1 billion transduced cells
2	3	55/F	None	Durvalumab, tremelimumab, sorafenib, FGRfr4 inhibitor	1.17 billion transduced cells
	4	57/M	Cirrhosis, alcoholic liver disease	Atezolizumab, bevacizumab	1.15 billion transduced cells
	5	41/F	None	Sorafenib, lenvatinib	1.14 billion transduced cells

Figure 4. ADP-A2AFP T-cell infiltration

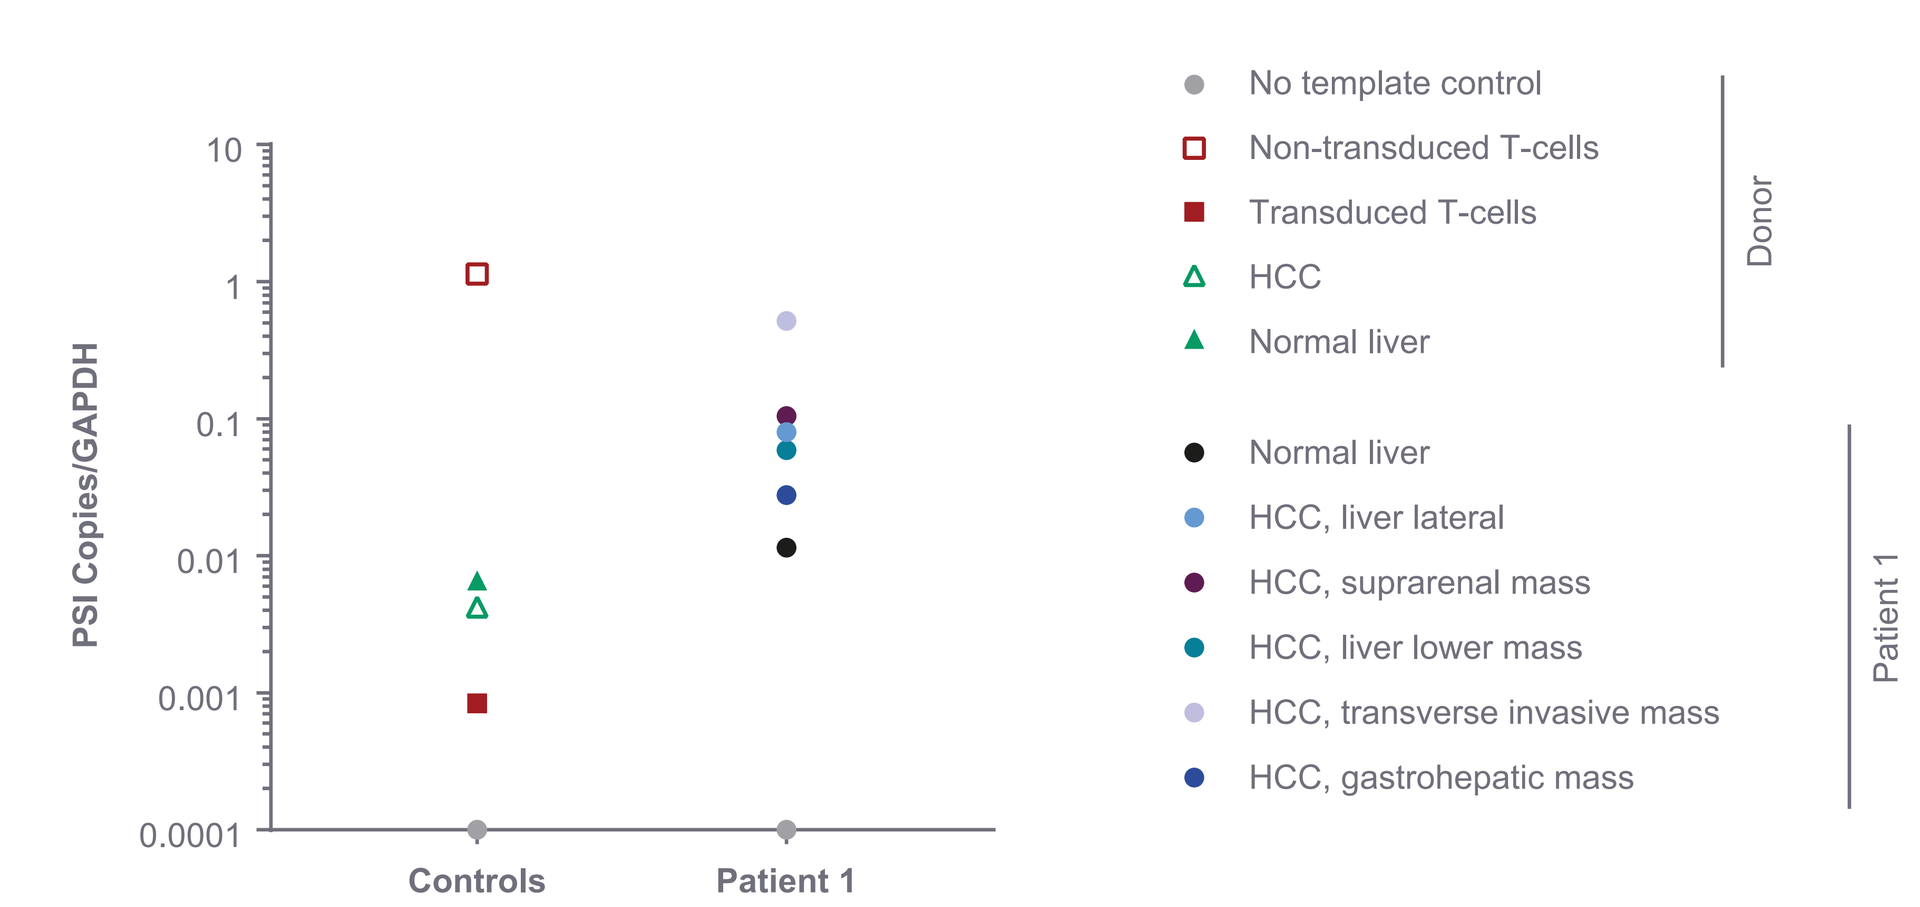
PSI Copies/GAPDH
10
1
0.1
0.01
0.001
0.0001
Controls
Patient 1
No template control
Non-transduced T-cells
Transduced T-cells
HCC
Normal liver
Donor
Normal liver
HCC, liver lateral
HCC, suprarenal mass
HCC, liver lower mass
HCC, transverse invasive mass
HCC, gastrohepatic mass
Patient 1

- Presence of ADP-A2AFP T-cells in post-treatment tumor samples was detected by digital PCR with primers specific to the PSI element
- Tumor tissue from a patient in Cohort 1 was acquired from 5 different lesions at autopsy 23 weeks after infusion (shown above)
- ADP-A2AFP T-cells were also detected in post-infusion tumor tissue from a patient treated in Cohort 2

Figure 5. Target lesion size following treatment

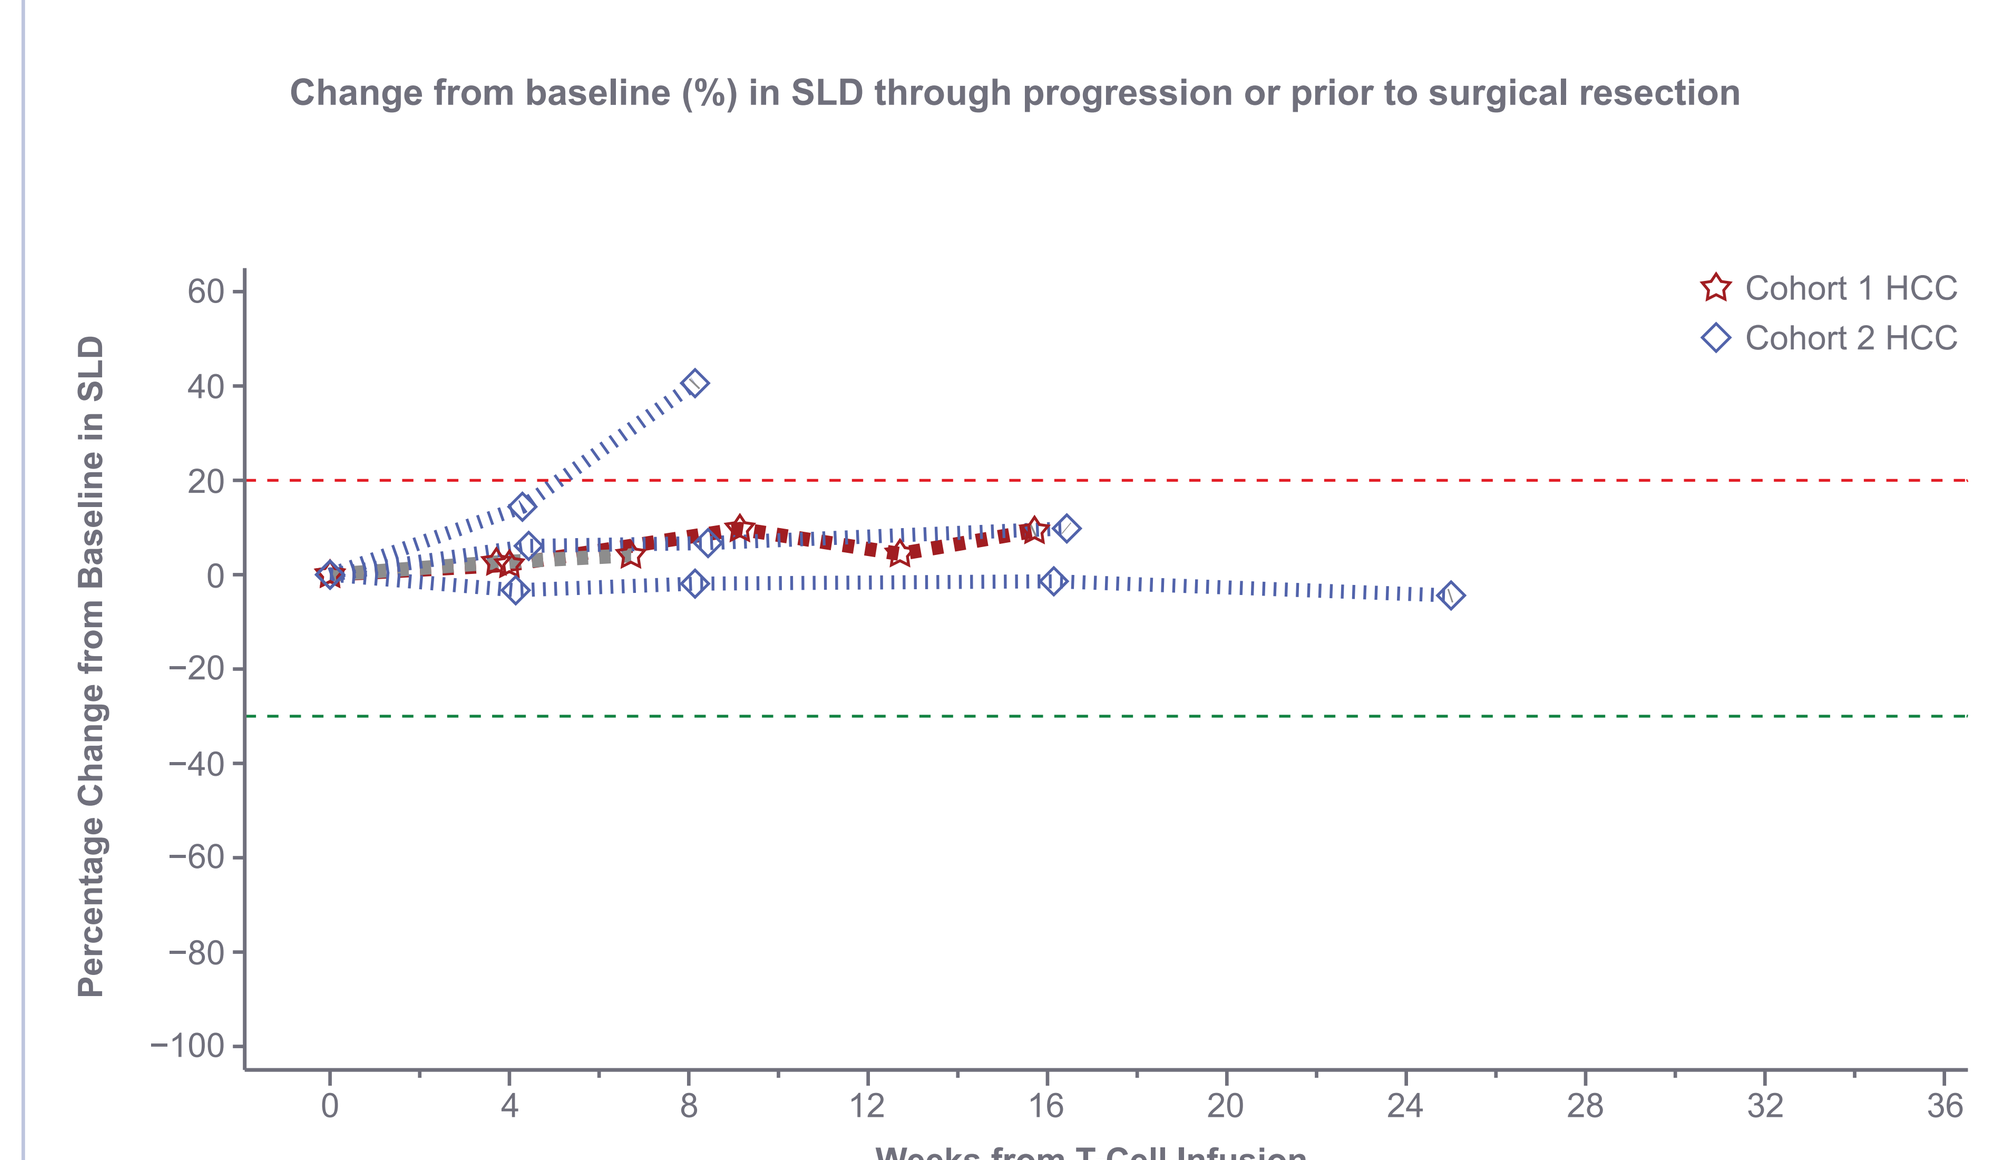
Change from baseline (%) in SLD through progression or prior to surgical resection
Cohort 1 HCC
Cohort 2 HCC
Percentage Change from Baseline in SLD
60
40
20
0
−20
−40
−60
−80
−100
0
4
8
12
16
20
24
28
32
36
Weeks from T-Cell Infusion

mITT: all patients who have met eligibility criteria and received at least 1 T-cell infusion
Cohort 1: 0.1 × 10^9 transduced cells, Cohort 2: 1.14–1.17 × 10^9 transduced cells

- Best overall response by RECISTv1.1 was stable disease for all 5 patients

Figure 6. Patient 3 (Cohort 2) baseline imaging and treatment results

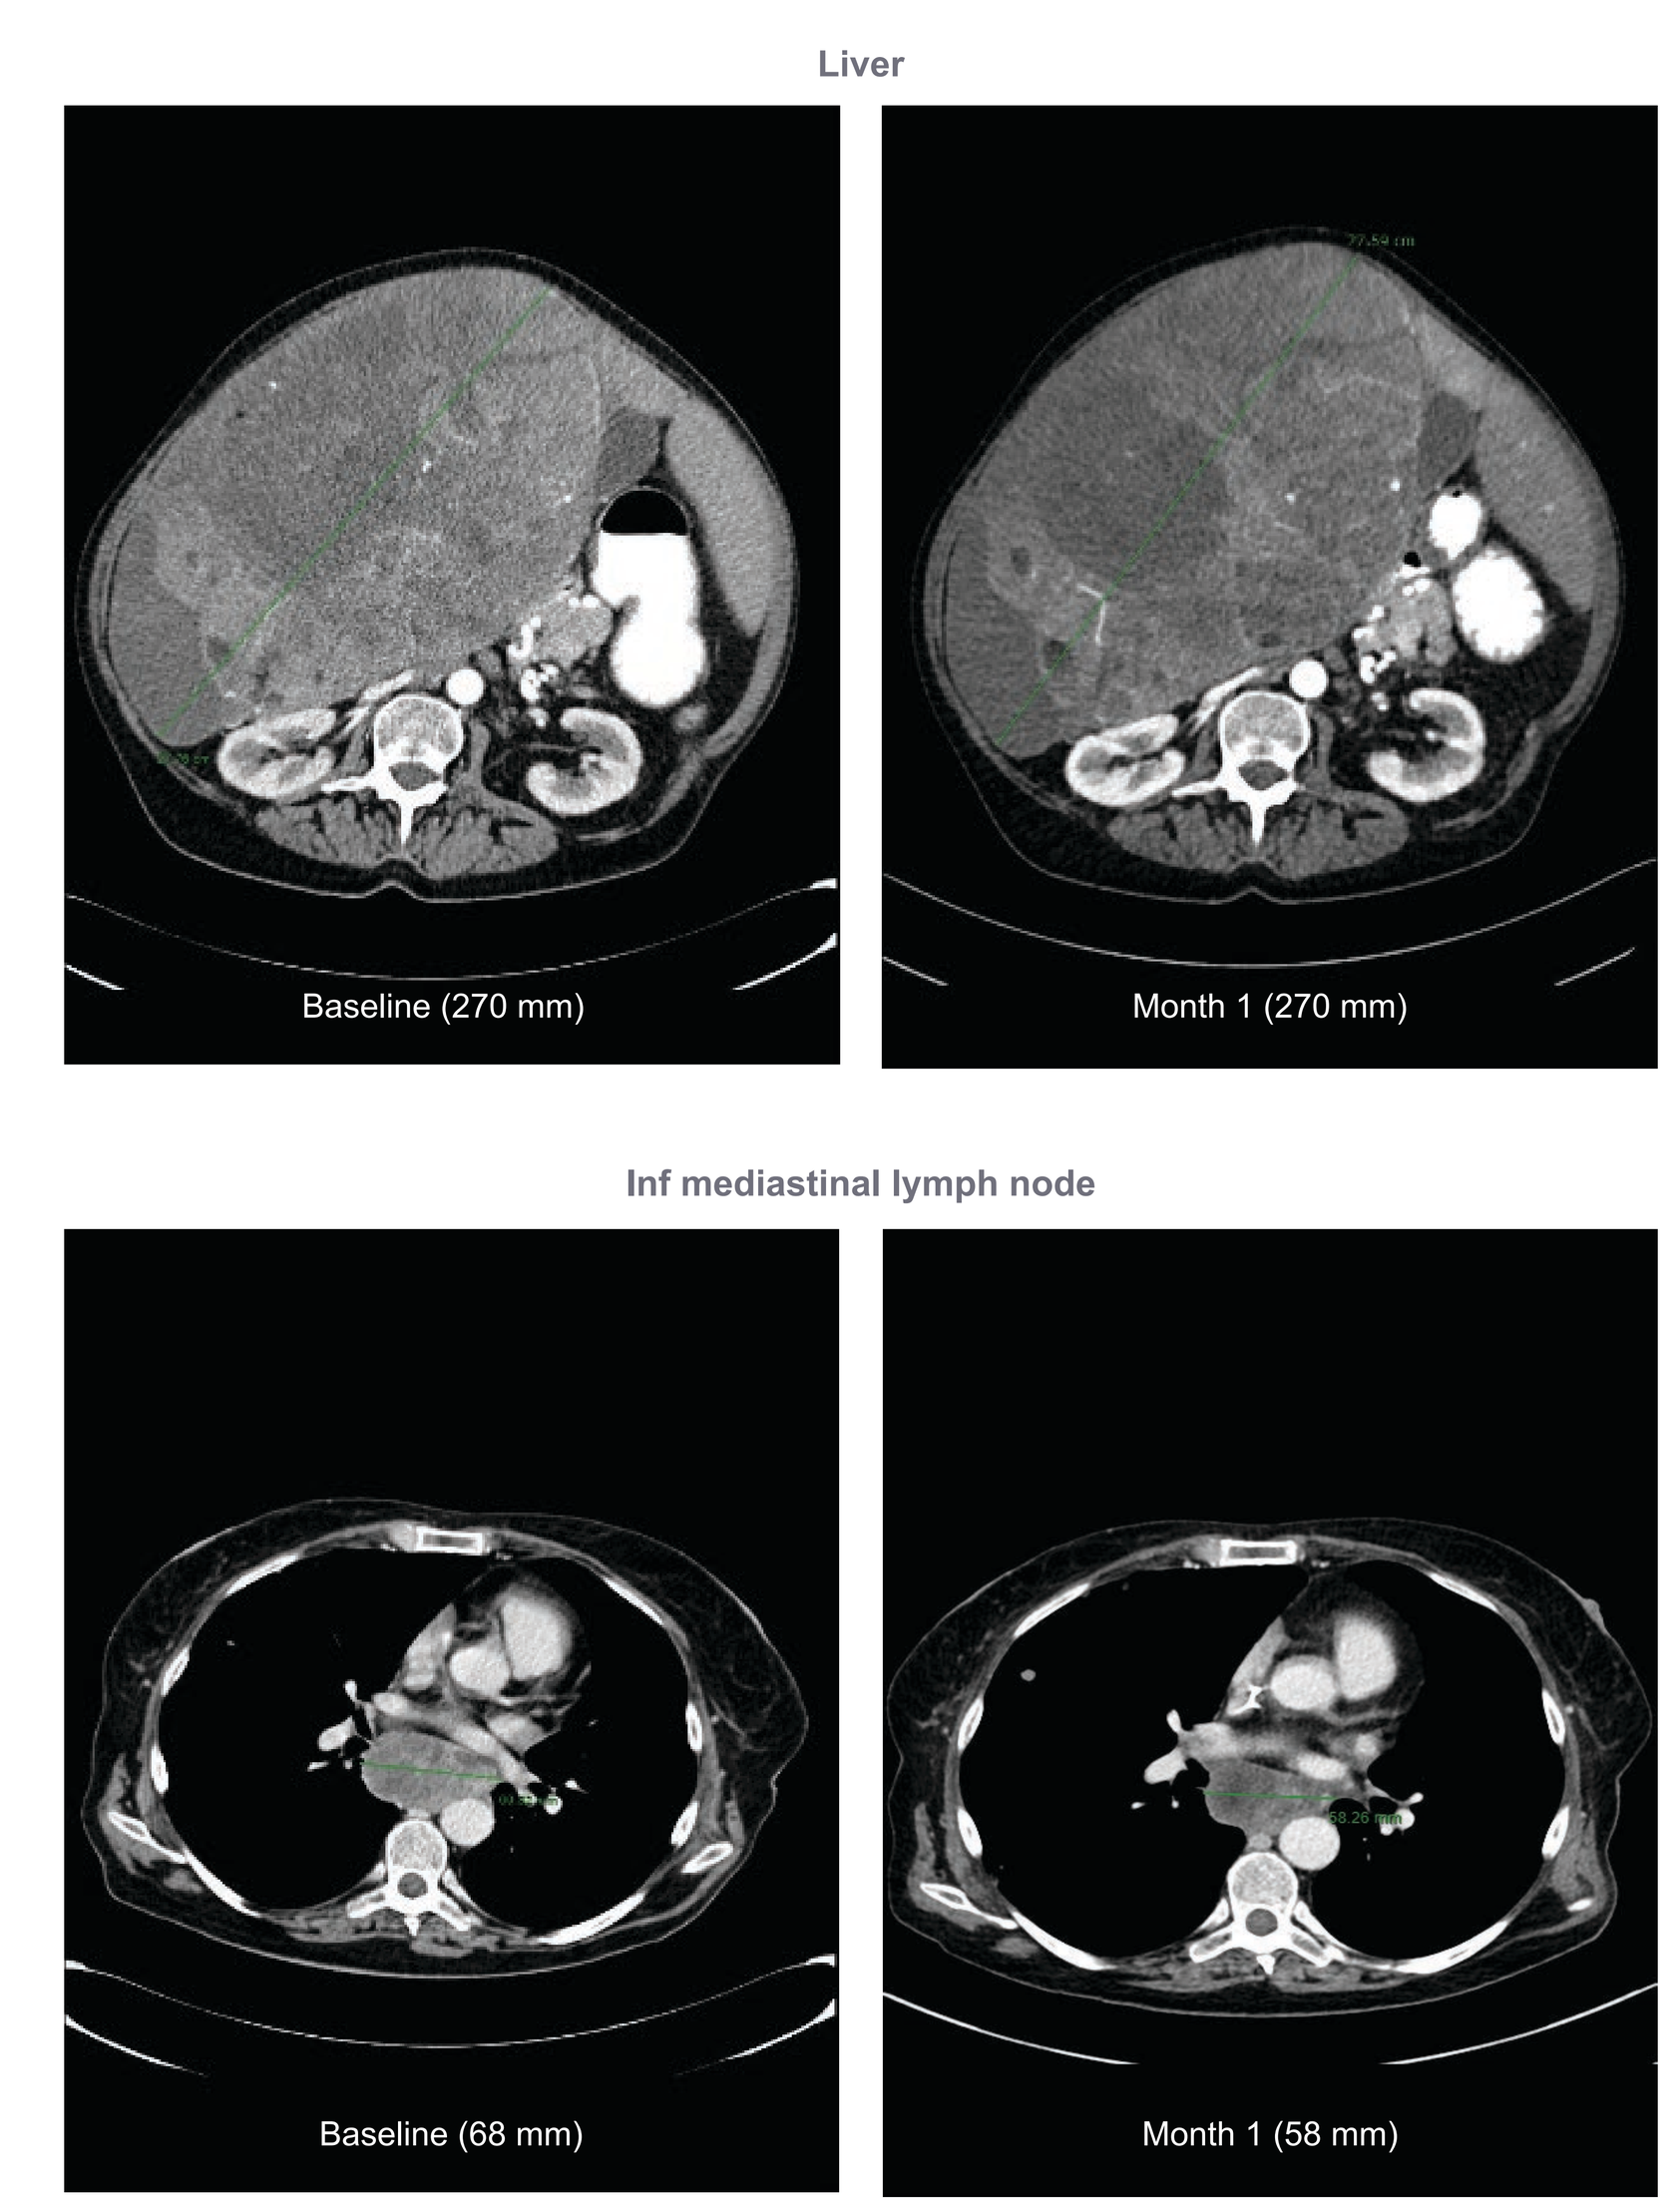
Liver
Baseline (270 mm)
Month 1 (270 mm)
Inf mediastinal lymph node
Baseline (68 mm)
Month 1 (58 mm)

- Large liver lesion unchanged and decrease in size of mediastinal lymph nodes at 1-month scan

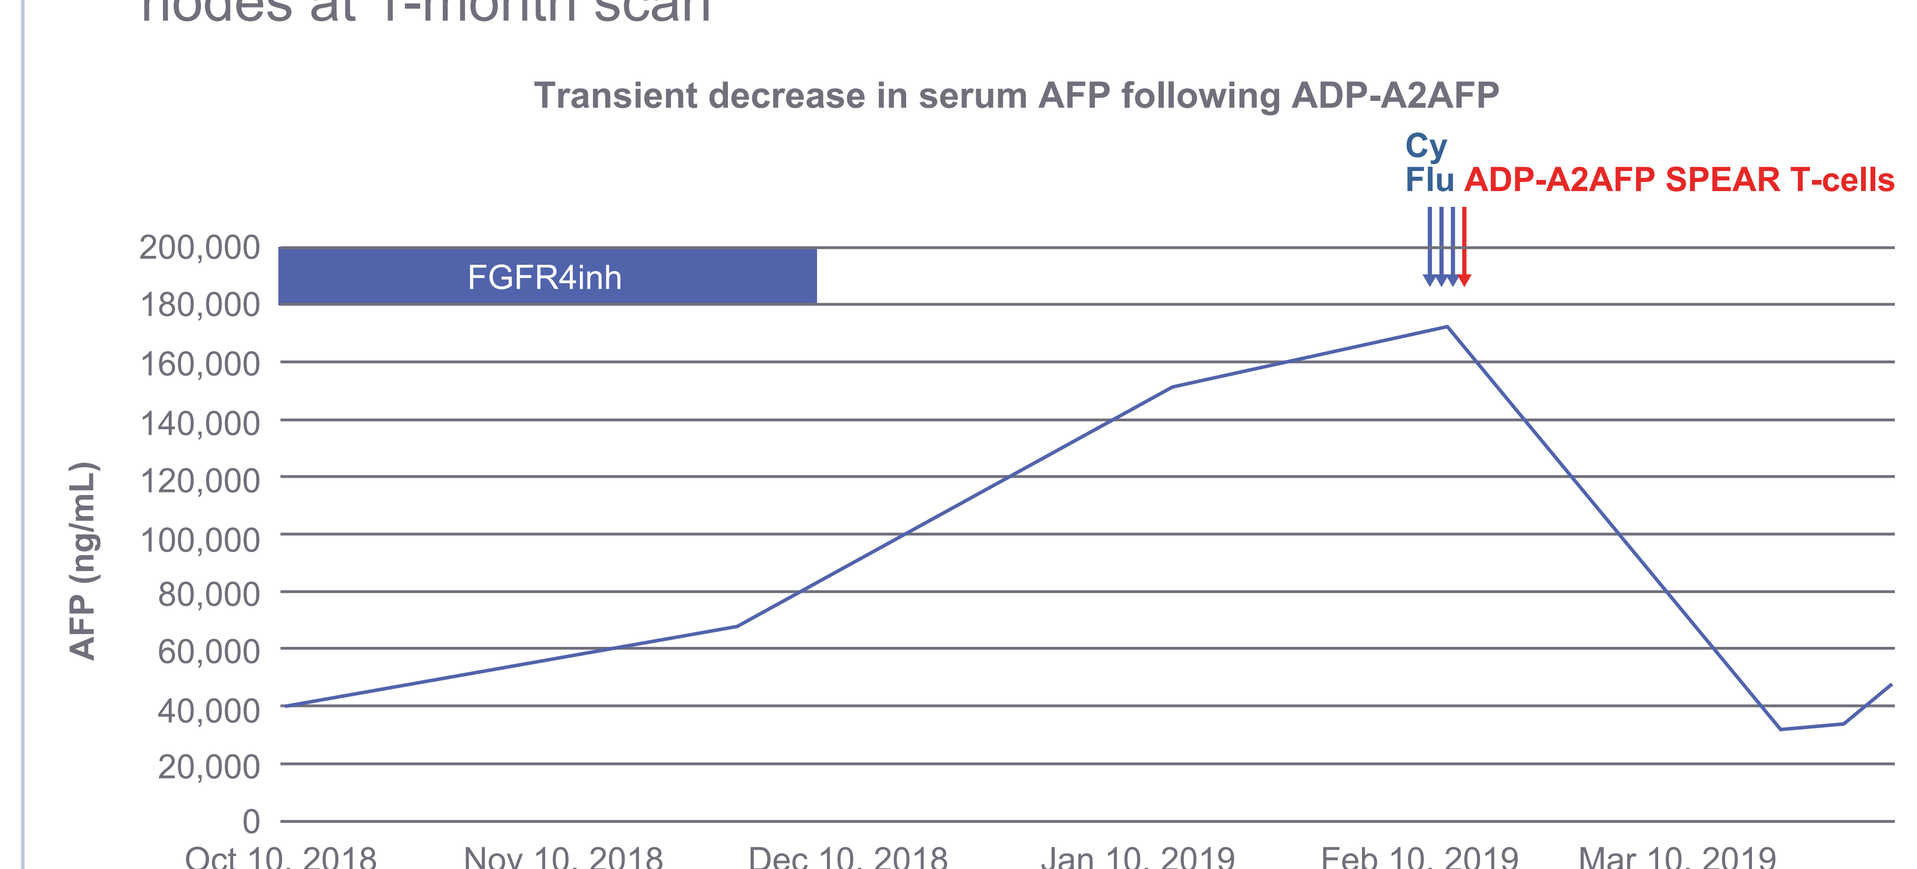
Transient decrease in serum AFP following ADP-A2AFP
Cy
Flu
ADP-A2AFP SPEAR T-cells
FGFR4inh
AFP (ng/mL)
200,000
180,000
160,000
140,000
120,000
100,000
80,000
60,000
40,000
20,000
0
Oct 10, 2018
Nov 10, 2018
Dec 10, 2018
Jan 10, 2019
Feb 10, 2019
Mar 10, 2019

Table 3. Any adverse event occurring in >1 patient in Cohorts 1 and 2

Term	Any Grade[a] N (%)	Grade ≥3 N (%)	Related[b] Any Grade N (%)	Related[b] Grade ≥3 N (%)
Patients with any AEs	5 (100)	5 (100)	5 (100)	5 (100)
Anemia	5 (100)	3 (60)	4 (80)	2 (40)
Leukopenia	5 (100)	5 (100)	5 (100)	5 (100)
Lymphopenia	5 (100)	5 (100)	5 (100)	5 (100)
Neutropenia	3 (60)	5 (100)	5 (100)	5 (100)
Thrombocytopenia	3 (60)	2 (40)	3 (60)	2 (40)
Activated partial thromboplastin time prolonged	2 (40)	1 (20)	0	0
Alopecia	2 (40)	0	2 (40)	0
Diarrhea	2 (40)	0	1 (20)	0
Fatigue	2 (40)	0	0	0
Hyponatremia	2 (40)	2 (40)	0	0
Pyrexia	2 (40)	0	1 (20)	0
Vomiting	2 (40)	0	1 (20)	0

[a]There were no Grade 5 AEs
[b]Related based on investigator assessment
Cytokine release syndrome related any grade occurred in 1 patient; no patient experienced Grade ≥3
Data cut-off July 6, 2020

Conclusions

- ADP-A2AFP was associated with an acceptable safety profile with no evidence of hepatotoxicity
- There have been no clear reports of T-cell related on-target or off-target toxicity, and no protocol-defined DLTs
- T-cell expansion and persistence appear to be dose-dependent, and ADP-A2AFP SPEAR T-cells were detectable in peripheral blood up to several months post-infusion
- There were transient decreases in tumor size and serum AFP in some patients, but no responses by RECIST v1.1 in these lower dose cohorts
- The trial is currently enrolling patients in the expansion phase at a higher dose level for HCC, and these data will be included in a separate oral presentation by Dr. Bruno Sangro[5]
- These early data support continued investigation with ADP-A2AFP

Abbreviations

AE, adverse event; AFP, alpha-fetoprotein; Cy, cyclophosphamide; DLT, dose-limiting toxicity; ECOG, Eastern Cooperative Oncology Group; FGFR4inh, fibroblast growth factor receptor 4 inhibitor; Flu, fludarabine; GAPDH, glyceraldehyde 3-phosphate dehydrogenase; HCC, hepatocellular carcinoma; HCV, hepatitis C virus; HLA, human leukocyte antigen; IHC, immunohistochemistry; mITT, modified intent-to-treat, PBMC, peripheral blood mononuclear cell; PSI, packaging signal; qPCR, quantitative polymerase chain reaction; SLD, sum of the longest diameters of target lesions; SPEAR, specific peptide enhanced affinity receptor; SRC, Safety Review Committee; TCR, T-cell receptor; XRT, radiation (x-ray) therapy

Acknowledgments

We are grateful to the patients and families who participated in this study
We thank the caregiving teams at all the clinical trial sites
The authors of this poster meet all the criteria for authorship suggested by the International Committee of Medical Journal Editors
Editorial support and formatting assistance for this poster were provided by Debra Brocksmith, MB ChB, PhD, of Elevate Scientific Solutions, which was contracted and compensated by Adaptimmune for these services

European Association for the Study of the Liver, The Digital International Liver Congress™; 27–29 August, 2020
Presenter contact information: Tim Meyer, t.meyer@ucl.ac.uk

References

1. Mizejewski GJ. *Anticancer Ther*. 2002;2:709-35
2. Cancer of the Liver Italian Program (CLIP) investigators. *Hepatology*. 1998;28:751-55
3. Bray F, et al. *CA Cancer J Clin*. 2018;68:394-424
4. Goyal L, et al. Presented at AACR March 29–April 3, 2019, Atlanta, Georgia, USA. Poster 3183
5. Sangro B, et al. Presented at EASL/ILC Digital Congress on 28 August, 2020. Oral presentation LBO12

Click the link or scan the QR code here to view Dr. Bruno Sangro's slides https://bit.ly/3g963hL

Full trial details from ClinicalTrials.gov can be viewed by scanning the QR code here https://clinicaltrials.gov/ct2/show/NCT03132792

SPEAR T-cell mechanism of action video can be viewed by scanning the QR code here https://youtu.be/zdI8IGXoQd0