Phase 1 Clinical Trial Evaluating the Safety of ADP-A2M10 SPEAR T-Cells in Patients with MAGE-A10⁺ Head and Neck, Melanoma, or Urothelial Tumors

David S. Hong[1], Marcus O. Butler[2], Russell Pachynski[3], Ryan J. Sullivan[4], Partow Kebriaei[1], Sarah Boross-Harmer[2], Matthew Frigault[5], Ecaterina Ileana Dumbrava[1], Amy Sauer[6], Francine Brophy[6], Jean-Marc Navenot[6], Svetlana Fayngerts[6], Jane Bai[6], Elliot Norry[6], Paula M. Fracasso[6]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Princess Margaret Cancer Centre, Toronto, Ontario, Canada; [3]Washington University School of Medicine, St. Louis, MO, USA; [4]Massachusetts General Hospital, Harvard Medical School, Boston, MA, USA; [5]Massachusetts General Hospital, Boston, MA, USA; [6]Adaptimmune, Philadelphia, PA, USA

An electronic copy of the poster can be viewed by scanning the QR code here https://bit.ly/2SEVn0f

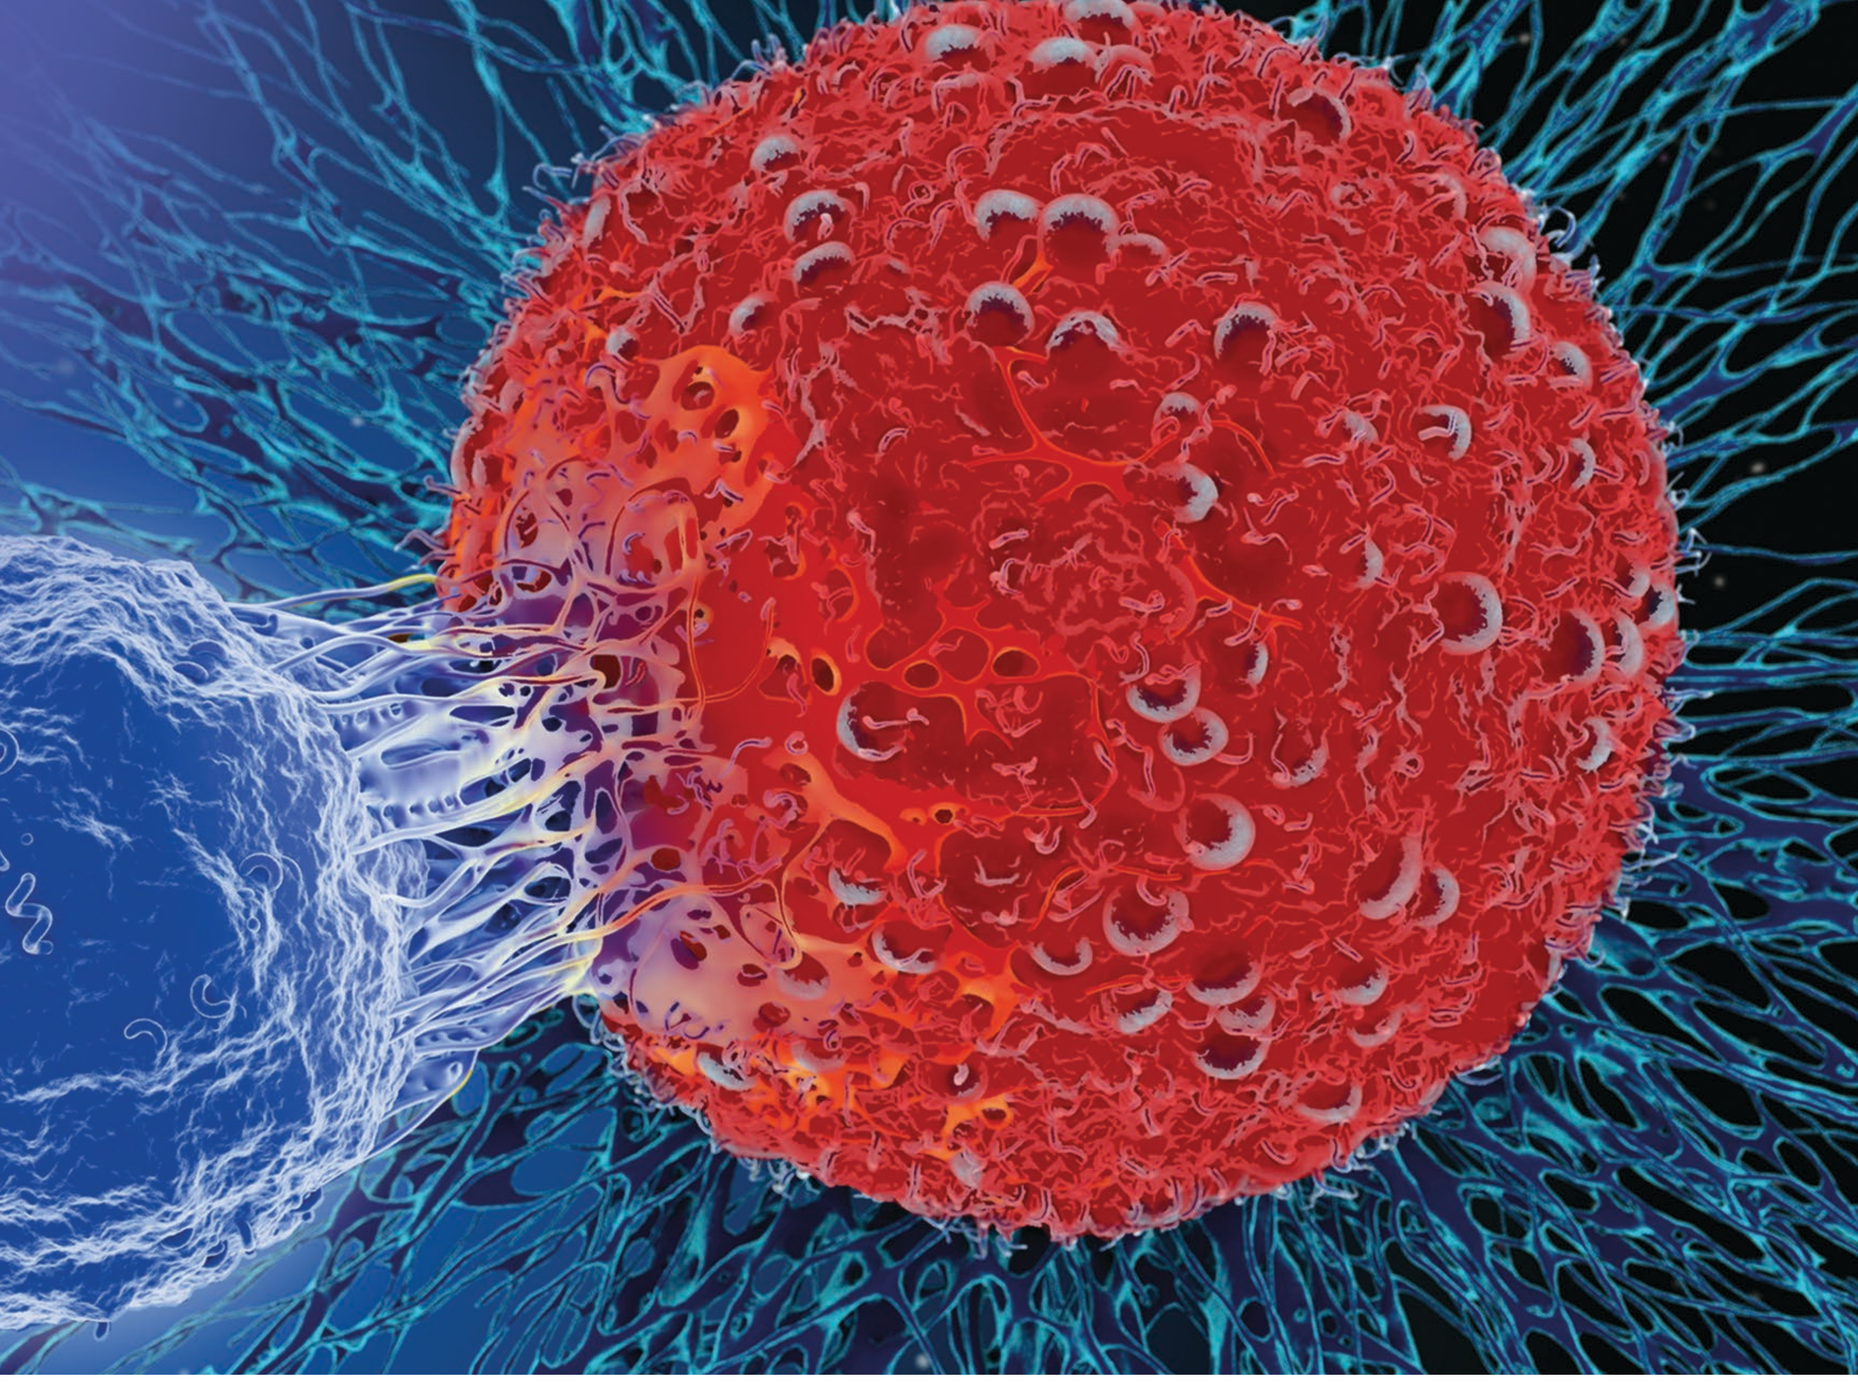

Background

- MAGE-A10 is expressed in 10%–50% of urothelial, melanoma, and head and neck cancers[1–3]
- ADP-A2M10 SPEAR T-cells are genetically engineered autologous T-cells that express a high affinity, MAGE-A10-specific TCR targeting MAGE-A10⁺ tumors in the context of HLA-A*02 (**Figure 1**)
- NCT02989064 was a first-in-human dose escalation trial that utilized a modified 3+3 design to evaluate safety and anti-tumor activity

Figure 1. SPEAR T-Cells Targeting MAGE-A10

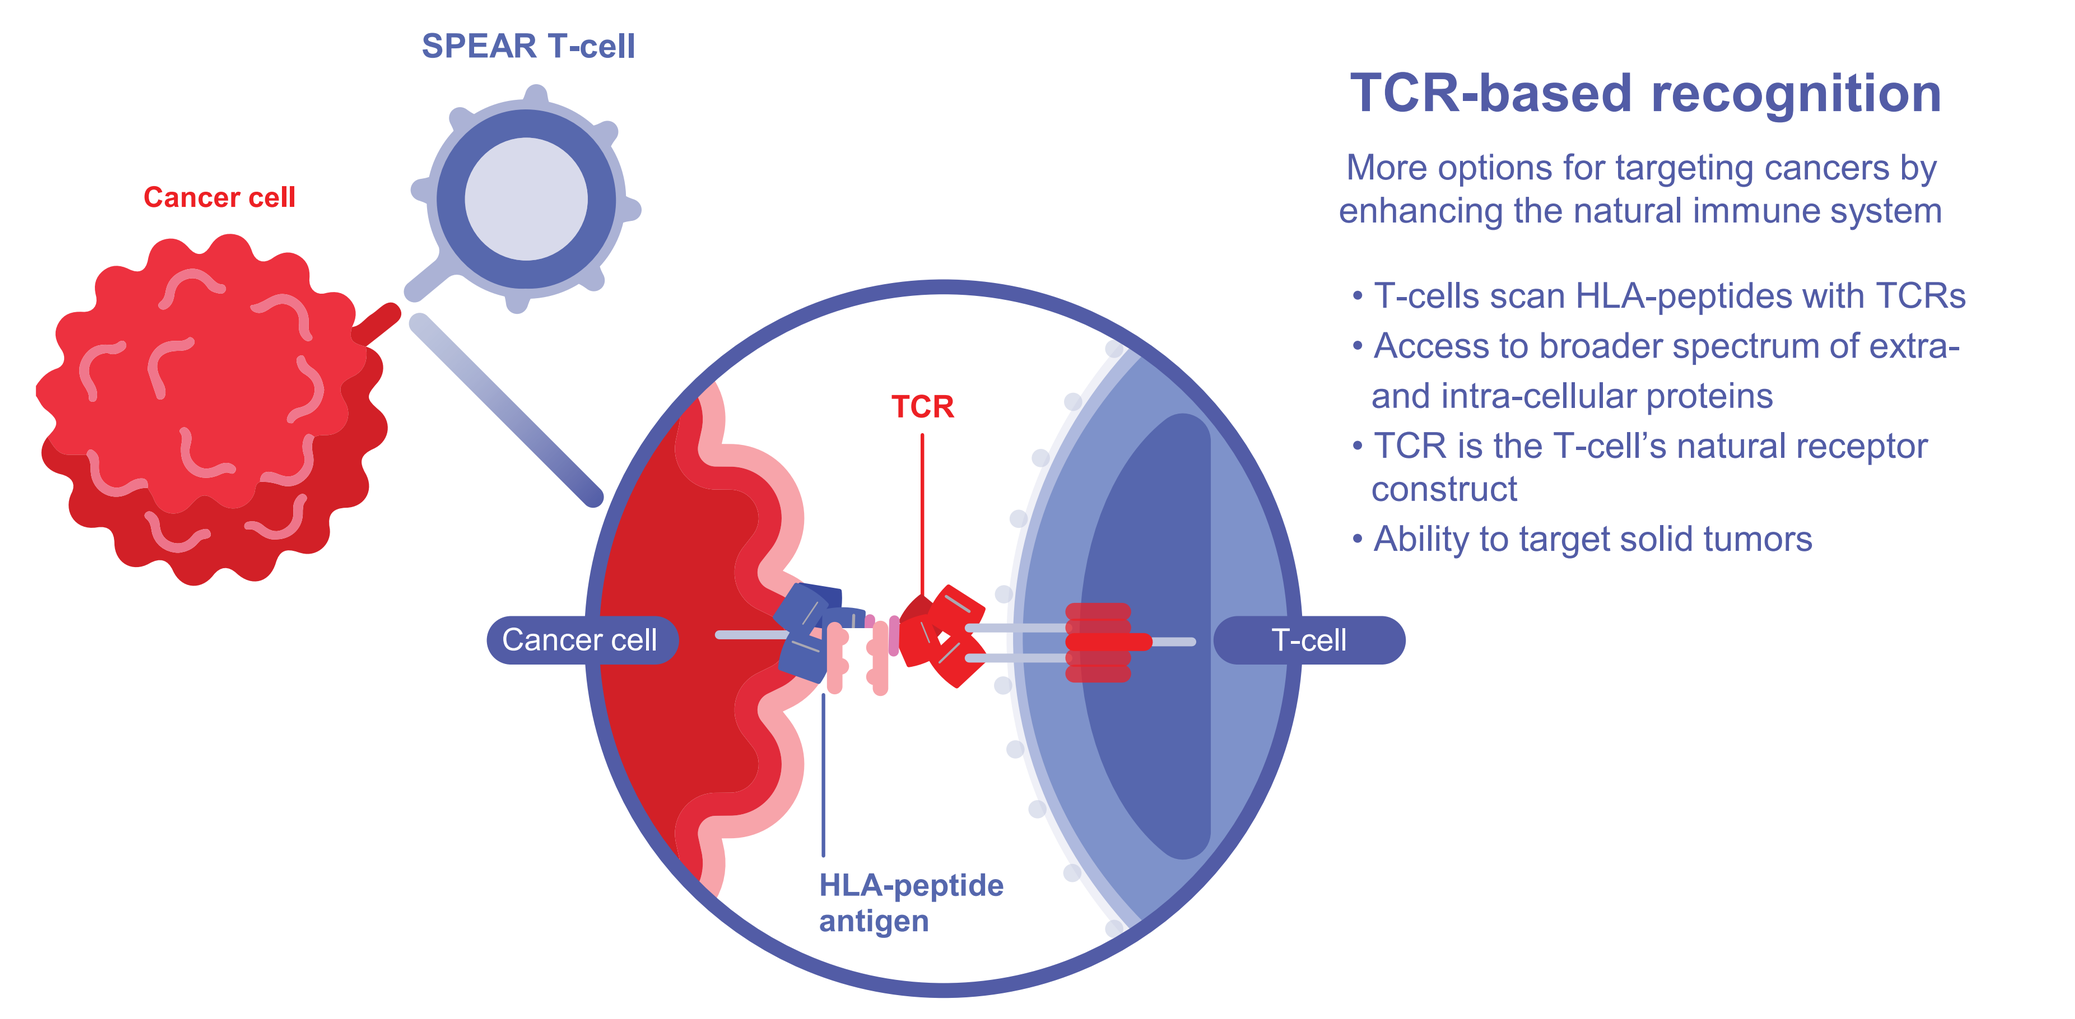
SPEAR T-cell
Cancer cell
TCR
Cancer cell
T-cell
HLA-peptide antigen
TCR-based recognition
More options for targeting cancers by enhancing the natural immune system
• T-cells scan HLA-peptides with TCRs
• Access to broader spectrum of extra- and intra-cellular proteins
• TCR is the T-cell's natural receptor construct
• Ability to target solid tumors

Objectives

Primary	• Evaluate the safety and tolerability of ADP-A2M10
Secondary	• Evaluate the anti-tumor activity of ADP-A2M10
Exploratory	• Identify serum and tumor factors that influence response or resistance to ADP-A2M10

Key Eligibility Criteria

Inclusion Criteria

- HLA-A*02:01 and/or *02:06
- Tumor expressing MAGE-A10
- Inoperable or metastatic (advanced) urothelial cancer
 - Received at least one prior systemic therapy in the adjuvant or metastatic setting; may have received treatment with a PD-1 or PD-L1 inhibitor
- Inoperable or metastatic (advanced) melanoma
 - Received PD-1 inhibitor and/or a CTLA-4 inhibitor
 - Received a BRAF inhibitor or combination of BRAF and MEK inhibitors for BRAFv600 mutant melanoma
- Inoperable or metastatic (advanced) squamous cell head and neck cancer
 - May have received or refused a platinum-containing chemotherapy for treatment of primary tumor in adjuvant or metastatic setting; may have received prior immunotherapy
- Defined washout periods for preceding anti-cancer therapy
- Measurable disease according to RECIST v1.1 prior to lymphodepletion
- ECOG status 0–1 and adequate organ function

Exclusion Criteria

- Patients with a history of symptomatic CNS metastases must have received treatment and be neurologically stable; patients with leptomeningeal disease or carcinomatous meningitis were not eligible
- Patients must not have active immune disorders requiring chronic steroid use

Trial Design

- Patients underwent lymphodepletion of fludarabine and cyclophosphamide prior to receiving ADP-A2M10
- ADP-A2M10 was administered at DL1, 0.1 × 10^9; DL2, >1.2–6 × 10^9; and DL3/Expansion, 1.2–15 × 10^9 transduced cells
- DLTs were determined regardless of attribution to cell infusion and adjudicated by a Safety Review Committee
- The interventional phase lasted until PD, death, or withdrawal, after which patients were rolled over into long-term follow-up for up to 15 years post-infusion
- ADP-A2M10 T-cell infiltration in post-treatment tumor tissue was assessed by duplex immunohistochemistry/RNAscope assay with antibody specific to CD3 and RNA probes specific to the ADP-A2M10 TCR vector construct

Figure 2. Trial Design

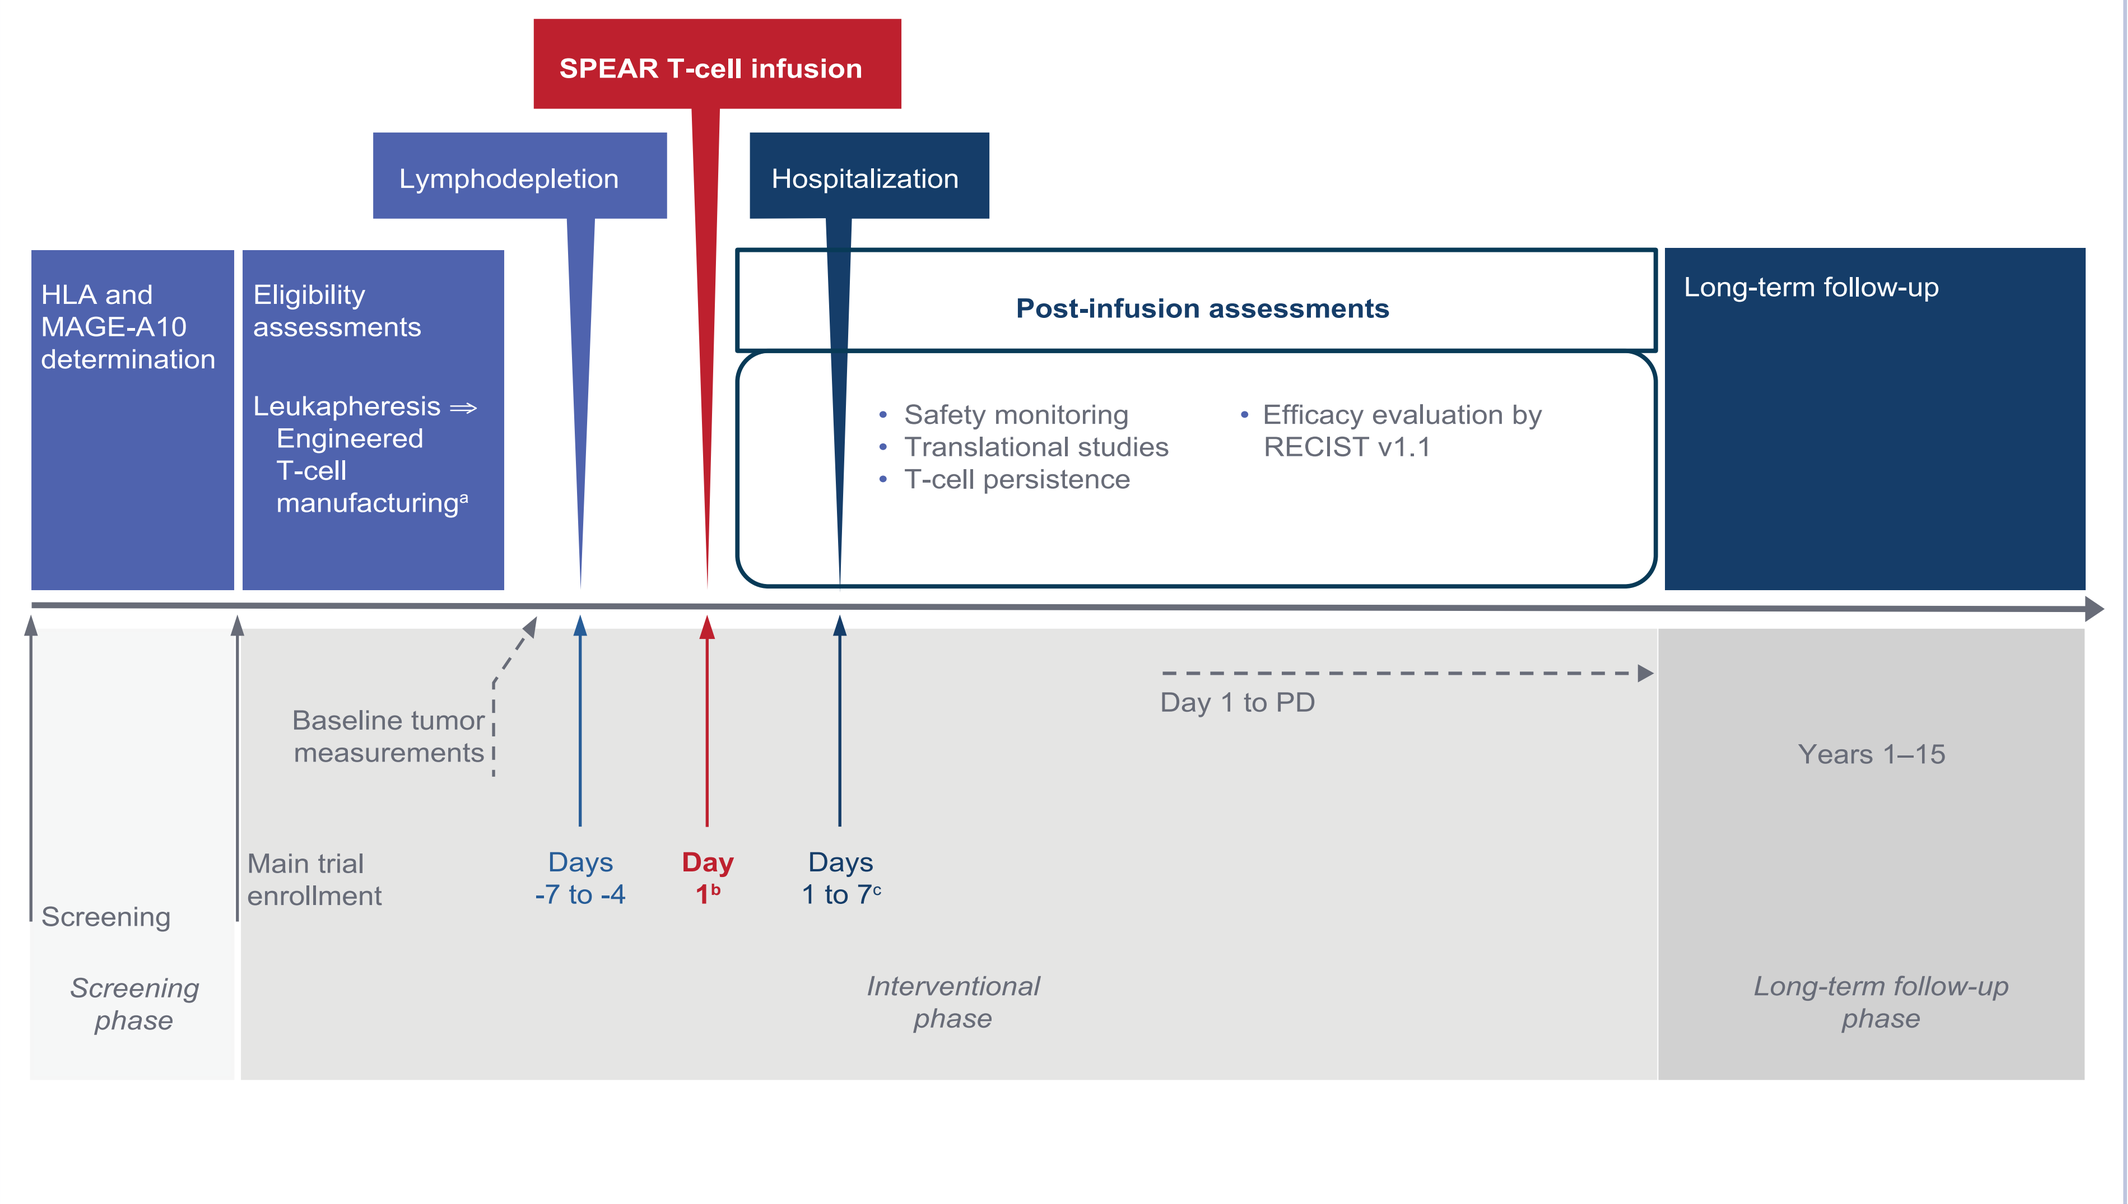
SPEAR T-cell infusion
Lymphodepletion
Hospitalization
HLA and MAGE-A10 determination
Eligibility assessments
Leukapheresis ⇒ Engineered T-cell manufacturing[a]
Post-infusion assessments
• Safety monitoring
• Translational studies
• T-cell persistence
• Efficacy evaluation by RECIST v1.1
Long-term follow-up
Baseline tumor measurements
Day 1 to PD
Years 1–15
Screening
Main trial enrollment
Days -7 to -4
Day 1[b]
Days 1 to 7[c]
Screening phase
Interventional phase
Long-term follow-up phase

[a]T-cell selection; lentiviral gene transfer of affinity-enhanced TCR; T-cell expansion
[b]Patient is hospitalized for T-cell infusion for a minimum of 3 days and discharged at the discretion of the investigator
[c]14 days in the UK

Figure 3. The Patient Cell Journey for Autologous SPEAR T-Cell Products

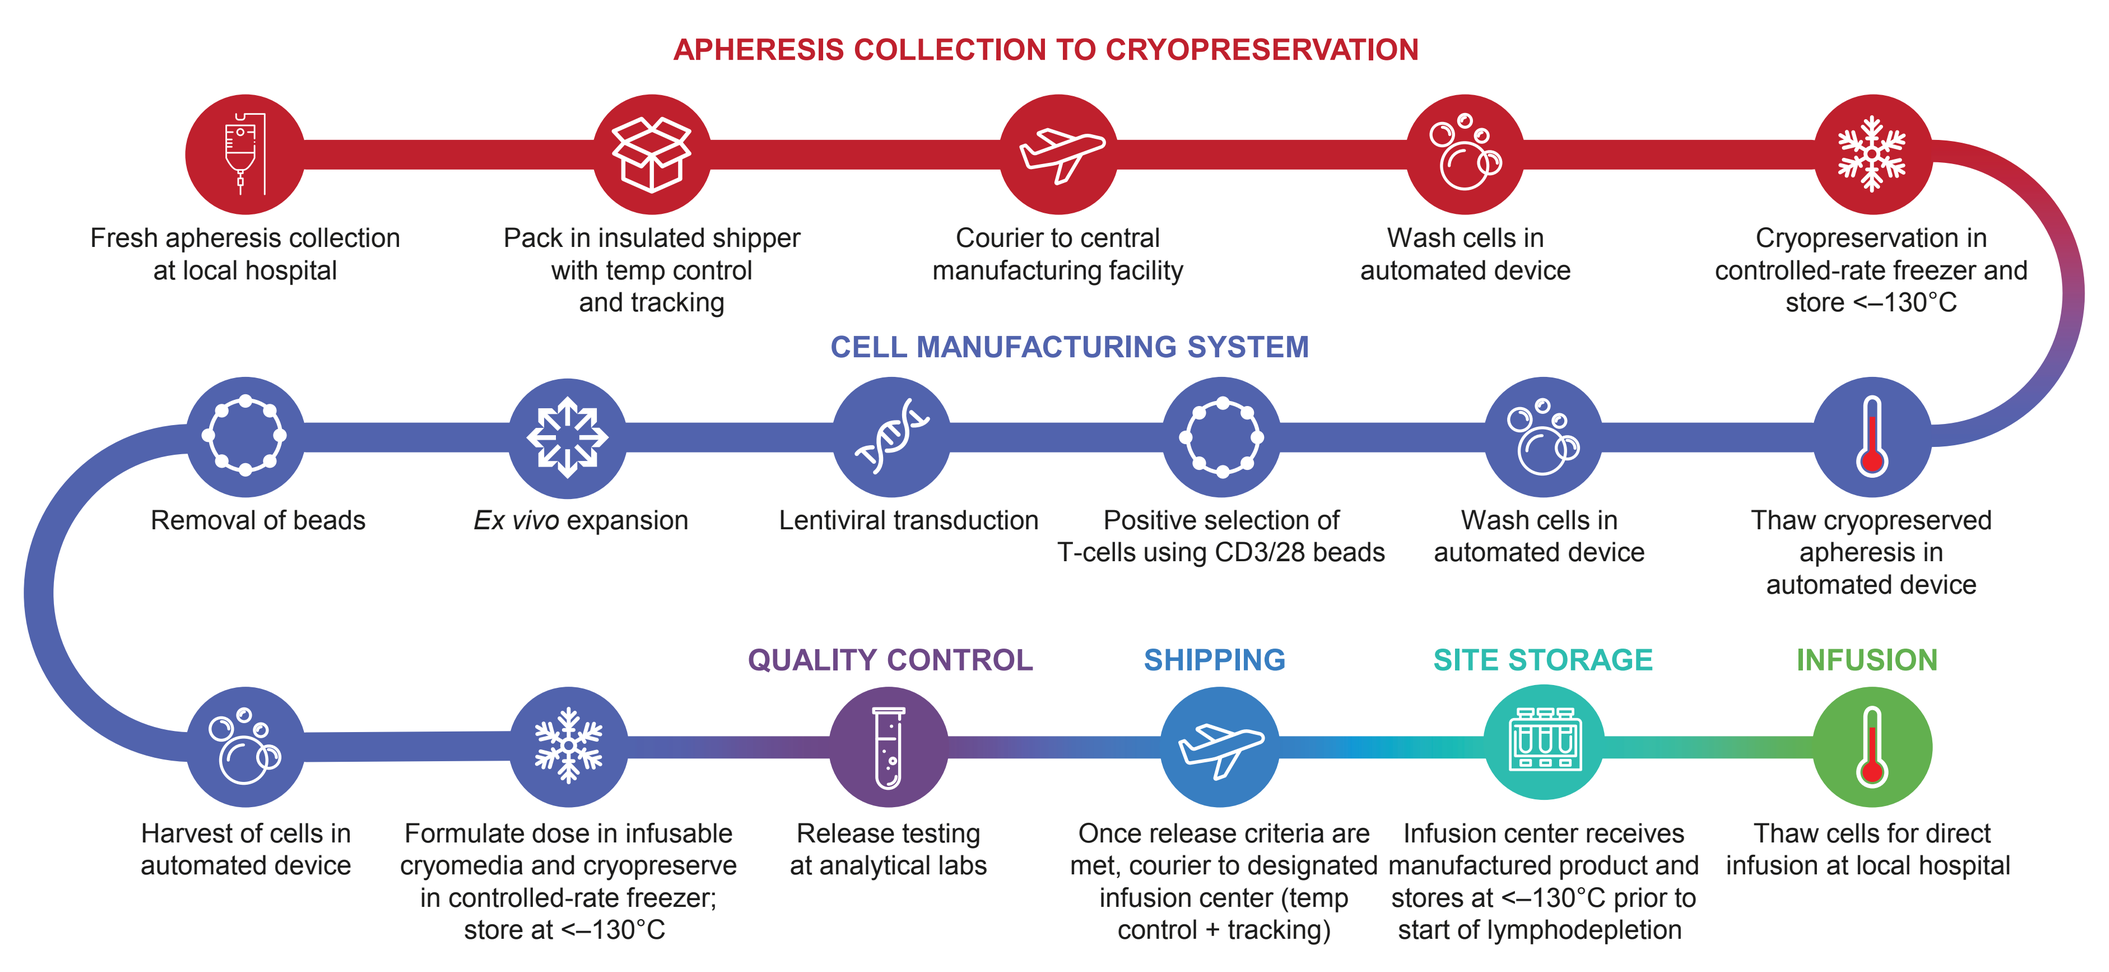
APHERESIS COLLECTION TO CRYOPRESERVATION
Fresh apheresis collection at local hospital
Pack in insulated shipper with temp control and tracking
Courier to central manufacturing facility
Wash cells in automated device
Cryopreservation in controlled-rate freezer and store <–130°C
CELL MANUFACTURING SYSTEM
Removal of beads
Ex vivo expansion
Lentiviral transduction
Positive selection of T-cells using CD3/28 beads
Wash cells in automated device
Thaw cryopreserved apheresis in automated device
QUALITY CONTROL
SHIPPING
SITE STORAGE
INFUSION
Harvest of cells in automated device
Formulate dose in infusable cryomedia and cryopreserve in controlled-rate freezer; store at <–130°C
Release testing at analytical labs
Once release criteria are met, courier to designated infusion center (temp control + tracking)
Infusion center receives manufactured product and stores at <–130°C prior to start of lymphodepletion
Thaw cells for direct infusion at local hospital

Results

- A total of 10 patients (8 male and 2 female) with head and neck (4), melanoma (3), and urothelial (3) cancers were treated (**Table 1**)
- Three patients were treated at DL1 and 7 patients were treated in DL3/Expansion
- The most frequently reported AEs ≥Grade 3 were lymphopenia, neutropenia, anemia, leukopenia, and thrombocytopenia, with one report of CRS Grade 3 (**Table 2**)
- SAEs are shown in **Table 3**
- Responses included: 4 patients, SD; 5 patients, PD; 1 patient unknown or missing (no post-baseline tumor assessment available) (**Table 4**)
- Peak ADP-A2M10 persistence appears to be dose dependent and trended higher in patients with SD (**Figure 4**)
- Transduced cells were detectable in tumors post-infusion (**Figure 2**)

Table 1. Patient Characteristics

Characteristic	N=10
Sex, n (%)	
Male	8 (80.0)
Female	2 (20.0)
Median age, years (range)	63 (46–76)
Race, n (%)	
White	9 (90)
Other	1 (10)
Tumor type, n (%)	
Head and neck cancer	4 (40)
Melanoma	3 (30)
Urothelial cancer	3 (30)
ECOG performance status, n (%)	
0	1 (10)
1	9 (90)

Table 2. Adverse Events in ≥25% of Patients by Frequency and Grade

N=10, n (%)	Any Grade	Grade 3	Grade 4
Patients with any AE	10 (100)	10 (100)	10 (100)
Leukopenia/WBC decreased	10 (100)	0	10 (100)
Lymphopenia/lymphocyte count decreased	10 (100)	0	10 (100)
Neutropenia/neutrophil count decreased	10 (100)	0	10 (100)
Anemia/RBC decreased	9 (90)	9 (90)	0
Fatigue	7 (70)	2 (20)	0
Hyponatremia	6 (60)	5 (50)	0
Pyrexia	6 (60)	1 (10)	0
Thrombocytopenia/platelet count decreased	6 (60)	1 (10)	4 (40)
Decreased appetite	5 (50)	1 (10)	0
Rash	5 (50)	0	1 (10)
Vomiting	5 (50)	0	0
Constipation	4 (40)	1 (10)	0
Diarrhea	4 (40)	0	0
Hypophosphatemia	4 (40)	1 (10)	1 (10)
Hypotension	4 (40)	1 (10)	0
Cough	3 (30)	0	0
Dehydration	3 (30)	0	0
Dizziness	3 (30)	0	0
Fall	3 (30)	0	0
Hypocalcemia	3 (30)	1 (10)	0
Hypomagnesemia	3 (30)	0	0
Nausea	3 (30)	0	0
Tumor pain	3 (30)	0	0

There were 2 reports of CRS. No grade 5 events were reported.

Table 3. Serious Adverse Events (All Doses)

N=10, n (%)	Any SAE	Related SAE[a]
Patients with any SAE	8 (80)	2 (20)
Acute kidney injury	2 (20)	1 (10)
Neutropenia/neutrophil count decreased	2 (20)	0
Abdominal pain	1 (10)	0
CRS	1 (10)	1 (10)
Failure to thrive	1 (10)	0
Hip fracture	1 (10)	0
Klebsiella sepsis	1 (10)	0
Pancytopenia	1 (10)	1 (10)
Rash	1 (10)	1 (10)
Sepsis	1 (10)	0
Small intestine obstruction	1 (10)	0
Thrombocytopenia/platelet count decreased	1 (10)	0
Tumor pain	1 (10)	0

[a]SAEs considered to be related to ADP-A2M10

Table 4. Best Overall Response for the First Infusion by Dose Group

	DL1 (n=3)	DL3 + Expansion (n=7)	All (n=10)
SD	0	4 (57)	4 (40)
PD	2 (67)	3 (43)	5 (50)
Unknown or missing	1 (33)	0	1 (10)

Figure 4. ADP-A2M10 T-Cells Persist in the Periphery for Several Months after Infusion

Peak persistence (copies/µg DNA)	DL1	DL3	Expansion	Overall
N	3	3	4	10
Mean	3398	97,828	70,899	58,727
Median	1265	82,549	64,160	55,643
Standard deviation	4653	31,762	58,322	54,394
Min; Max	194; 8735	76,592; 134,342	13,270; 142,004.2	194; 142,004

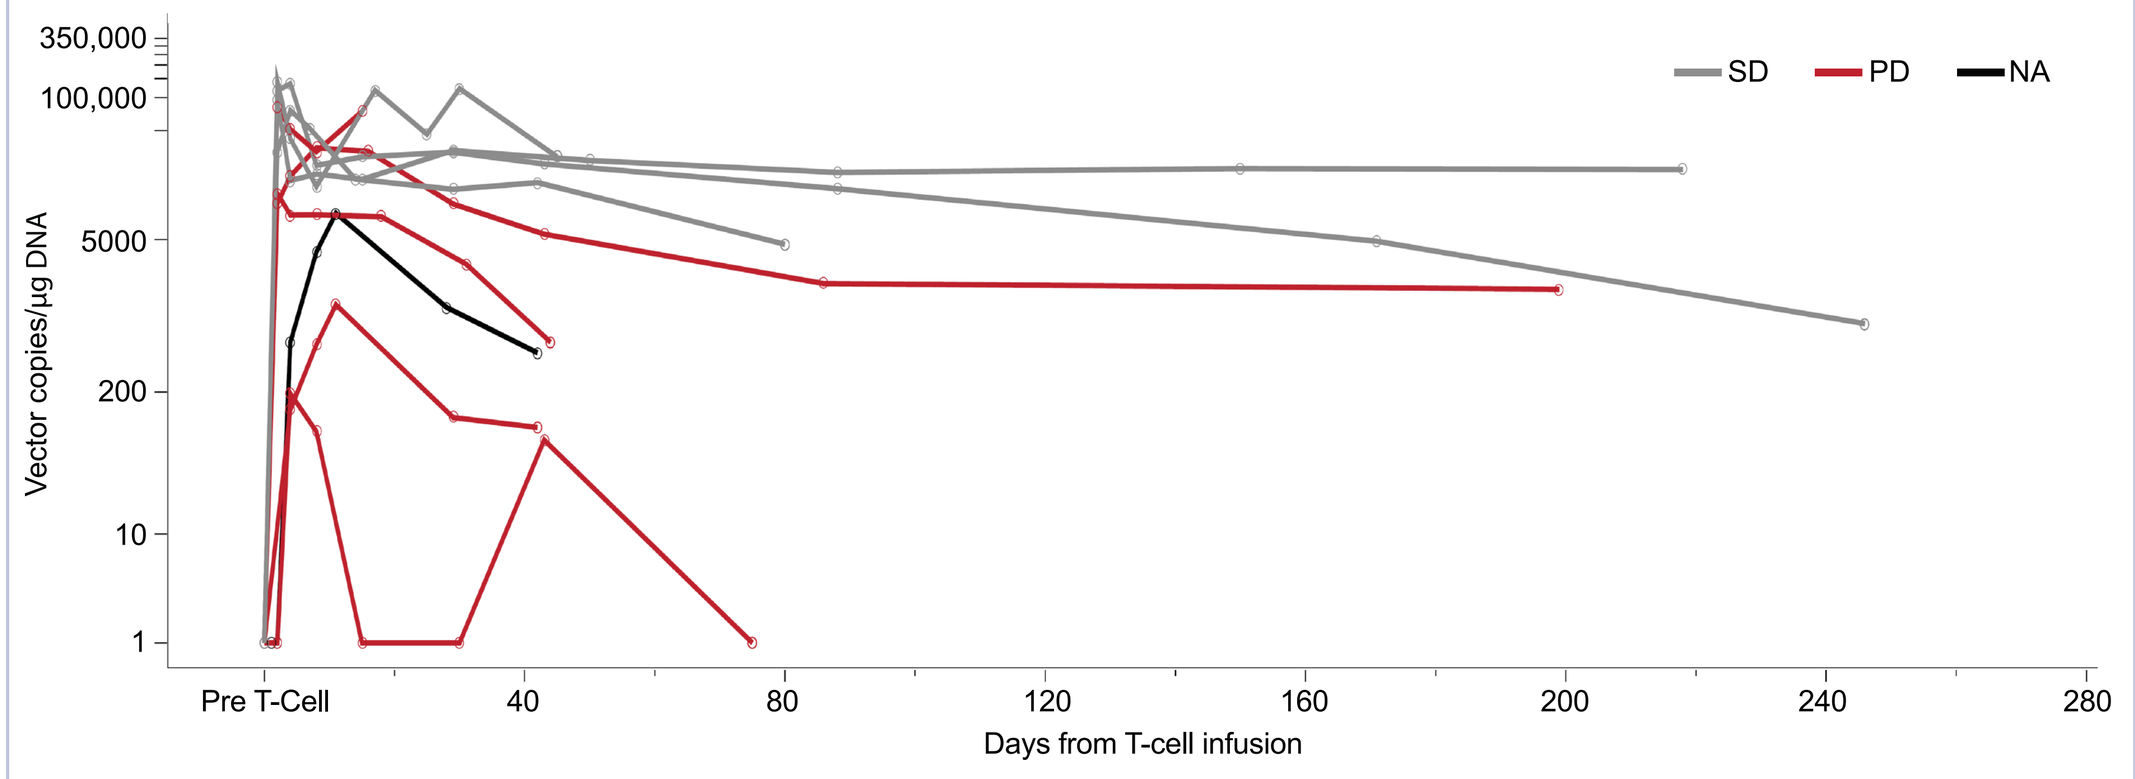
SD
PD
NA
350,000
100,000
5000
200
10
1
Vector copies/µg DNA
Pre T-Cell
40
80
120
160
200
240
280
Days from T-cell infusion

Persistence of ADP-A2M10 T-cells was measured by qPCR of the PSI element sequence in genomic DNA extracted from PBMCs

Figure 5. ADP-A2M10 T-Cells Were Detected in Two of Four Post-Infusion Tumor Samples

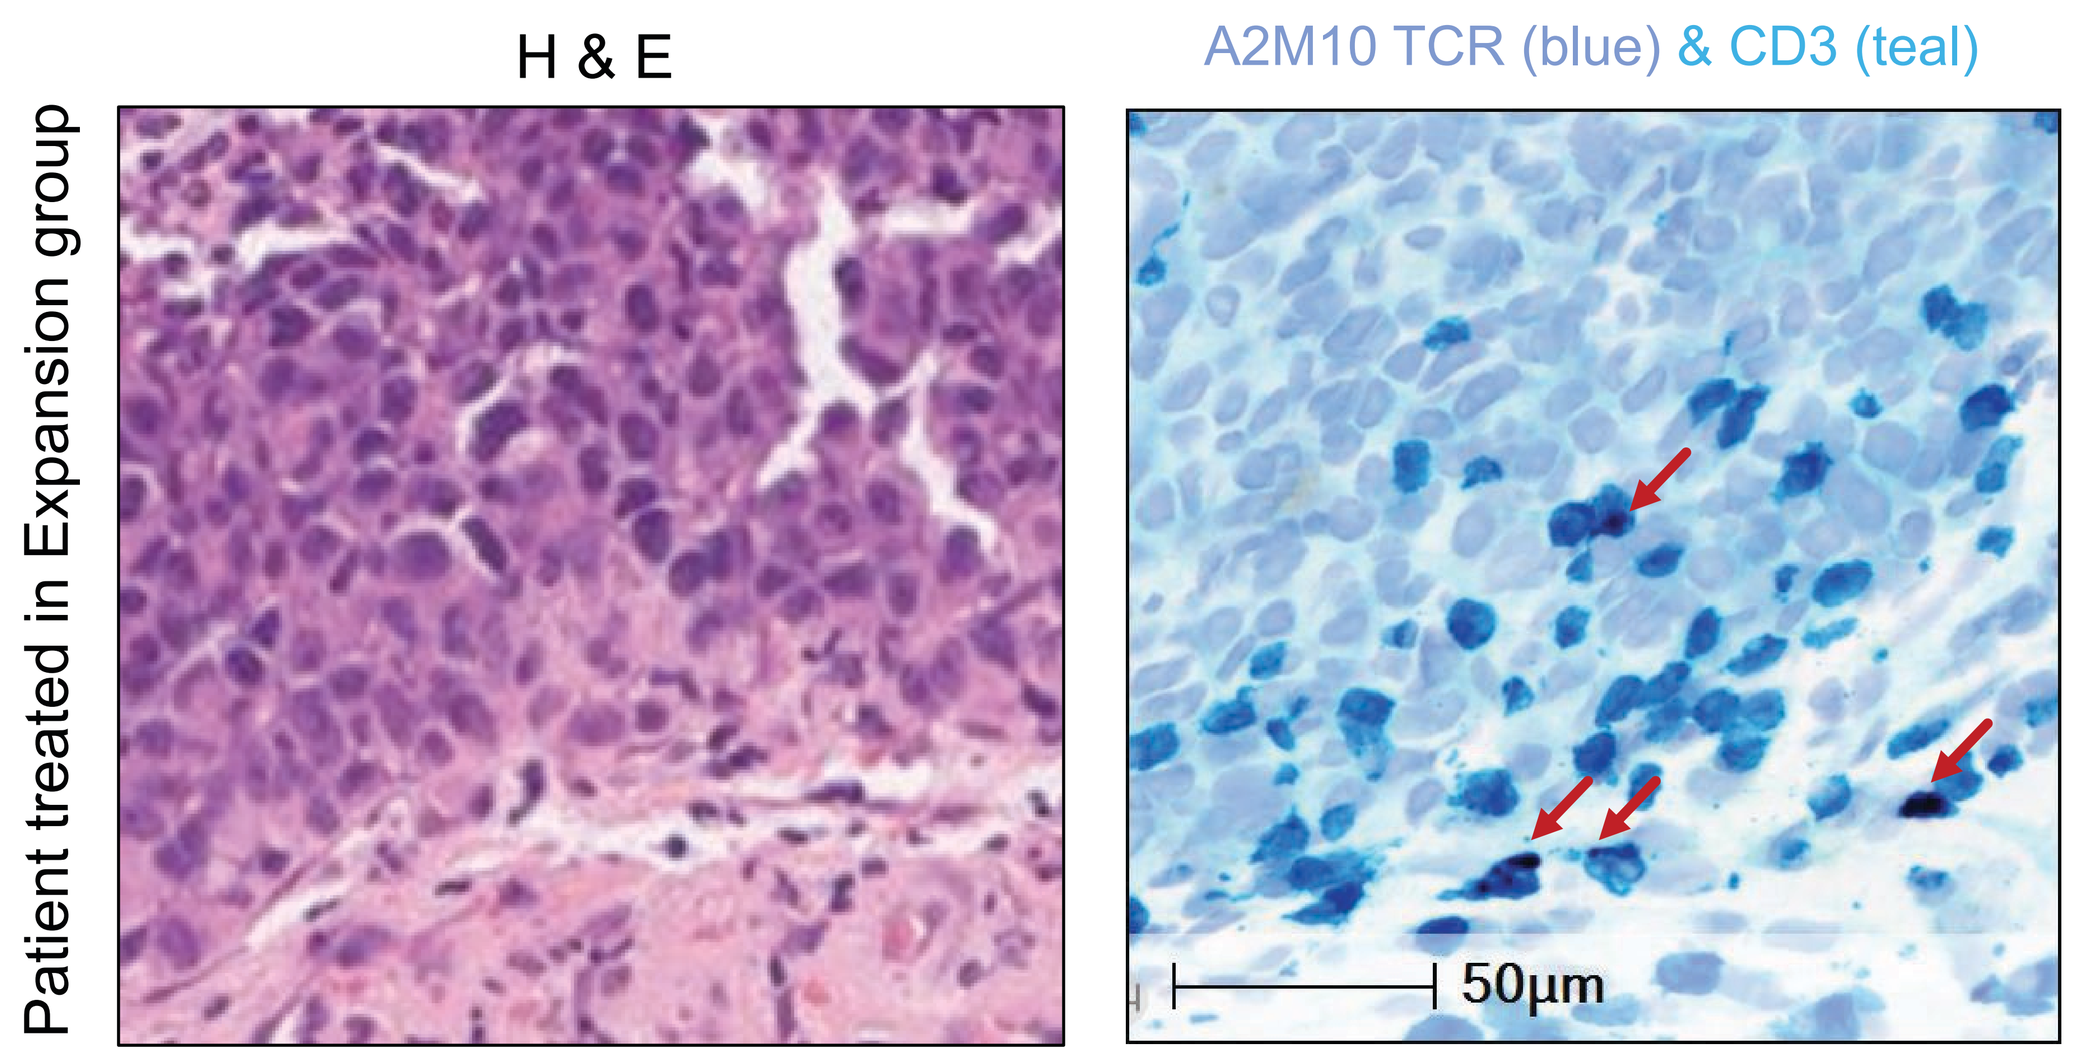
H & E
A2M10 TCR (blue) & CD3 (teal)
Patient treated in Expansion group
50µm

ADP-A2M10 T-cell infiltration was assessed by duplex immunohistochemistry/RNAscope assay with antibody specific to CD3 and RNA probes specific to the ADP-A2M10 TCR vector construct

Conclusions

- ADP-A2M10 SPEAR T-cells have shown acceptable safety and no evidence of toxicity related to off-target binding or alloreactivity
- Given the minimal anti-tumor activity and the discovery that MAGE-A10 expression frequently overlaps with MAGE-A4 expression, the clinical program has closed
- Several trials with SPEAR T-cells targeting MAGE-A4 are ongoing (https://bit.ly/35htsZK)

Abbreviations

AE, adverse event; CRS, cytokine release syndrome; CTLA-4, cytotoxic T-lymphocyte-associated protein 4; DL, dose level; DLT, dose-limiting toxicity; ECOG, Eastern Cooperative Oncology Group; HLA, human leukocyte antigen; MAGE-A10, melanoma-associated antigen-A10; NA, unknown or missing; PBMC, peripheral blood mononuclear cell; PD, progressive disease; PD-1, programmed cell death 1; PD-L1, programmed cell death ligand; PR, partial response; PSI, packaging signal; qPCR, quantitative PCR; RECIST, response evaluation criteria in solid tumors; SAE, serious adverse event; SD, stable disease; SPEAR, specific peptide enhanced affinity receptor; TCR, T-cell receptor

Acknowledgements and Disclosures

The authors would like to thank the patients and their caregivers for taking part in this trial, and the investigators and their teams who participated in this work

DS Hong: *Research/Grant Funding:* AbbVie, Adaptimmune, Aldi-Norte, Amgen, AstraZeneca, Bayer, BMS, Daiichi-Sankyo, Eisai, Fate Therapeutics, Genentech, Genmab, GSK, Ignyta, Infinity, Kite, Kyowa, Lilly, LOXO, Merck, MedImmune, Mirati, miRNA, Molecular Templates, Mologen, NCI-CTEP, Novartis, Pfizer, Seattle Genetics, Takeda, Turning Point Therapeutics; *Consulting or Advisory Role:* Adaptimmune, Alpha Insights, Amgen, Axiom, Baxter, Bayer, eCancer, Genentech, GLG, Group H, Guidepoint, Infinity, Libreum, Medscape, Numab, Oncology Education Project Association, Pfizer, Prime Oncology, Takeda, Trieza Therapeutics, WebMD; *Other Ownership Interests:* Molecular Match (Advisor), OncoResponse (Founder), Presagia Inc (Advisor); *Travel/Accommodations/Expenses:* Bayer, Genmab, AACR, ASCO, SITC

This trial (NCT02989064) was sponsored by Adaptimmune

Editorial support and formatting assistance were provided by Andy Shepherd, PhD, CMPP, of Elevate Scientific Solutions, which was contracted and compensated by Adaptimmune for these services

E-poster: Society for Immunotherapy of Cancer (SITC), Virtual Congress, November 9–14, 2020
Presenter contact information: David S. Hong, dshong@mdanderson.org

References

1. Daudi S, et al. *PLoS One*. 2014;9:e104099
2. Chunxia Su, et al. *Int J Clin Exp Pathol*. 2015;8:5509–18
3. Elke Schultz-Thater, et al. *Int J Cancer*. 2011;129:1137–48

Full trial details from ClinicalTrials.gov can be viewed by scanning the QR code here https://bit.ly/3iRWCUn

SPEAR T-cell mechanism of action video can be viewed by scanning the QR code here https://bit.ly/3huj9qa