Inhibition of AKT Signaling During Expansion of TCR-Engineered T-Cells from Patient Leukocyte Material Generates SPEAR T-Cells with Enhanced Functional Potential *In Vitro*

Katerina Mardilovich, Lili Wang, Rachel Kenneil, Gareth Betts, Natalie Bath, Will Spinner, Vanessa De Mello, Seint Lwin, Joseph P. Sanderson, Jonathan D. Silk, Alex Tipping, Andrew B. Gerry, Phil Bassett, Karen Miller, Mark Dudley, Emily M. Schmidt

Adaptimmune, Abingdon, Oxfordshire, UK, and Philadelphia, PA, USA

An electronic copy of the poster can be viewed by scanning the QR code here https://bit.ly/3jJ7CVq

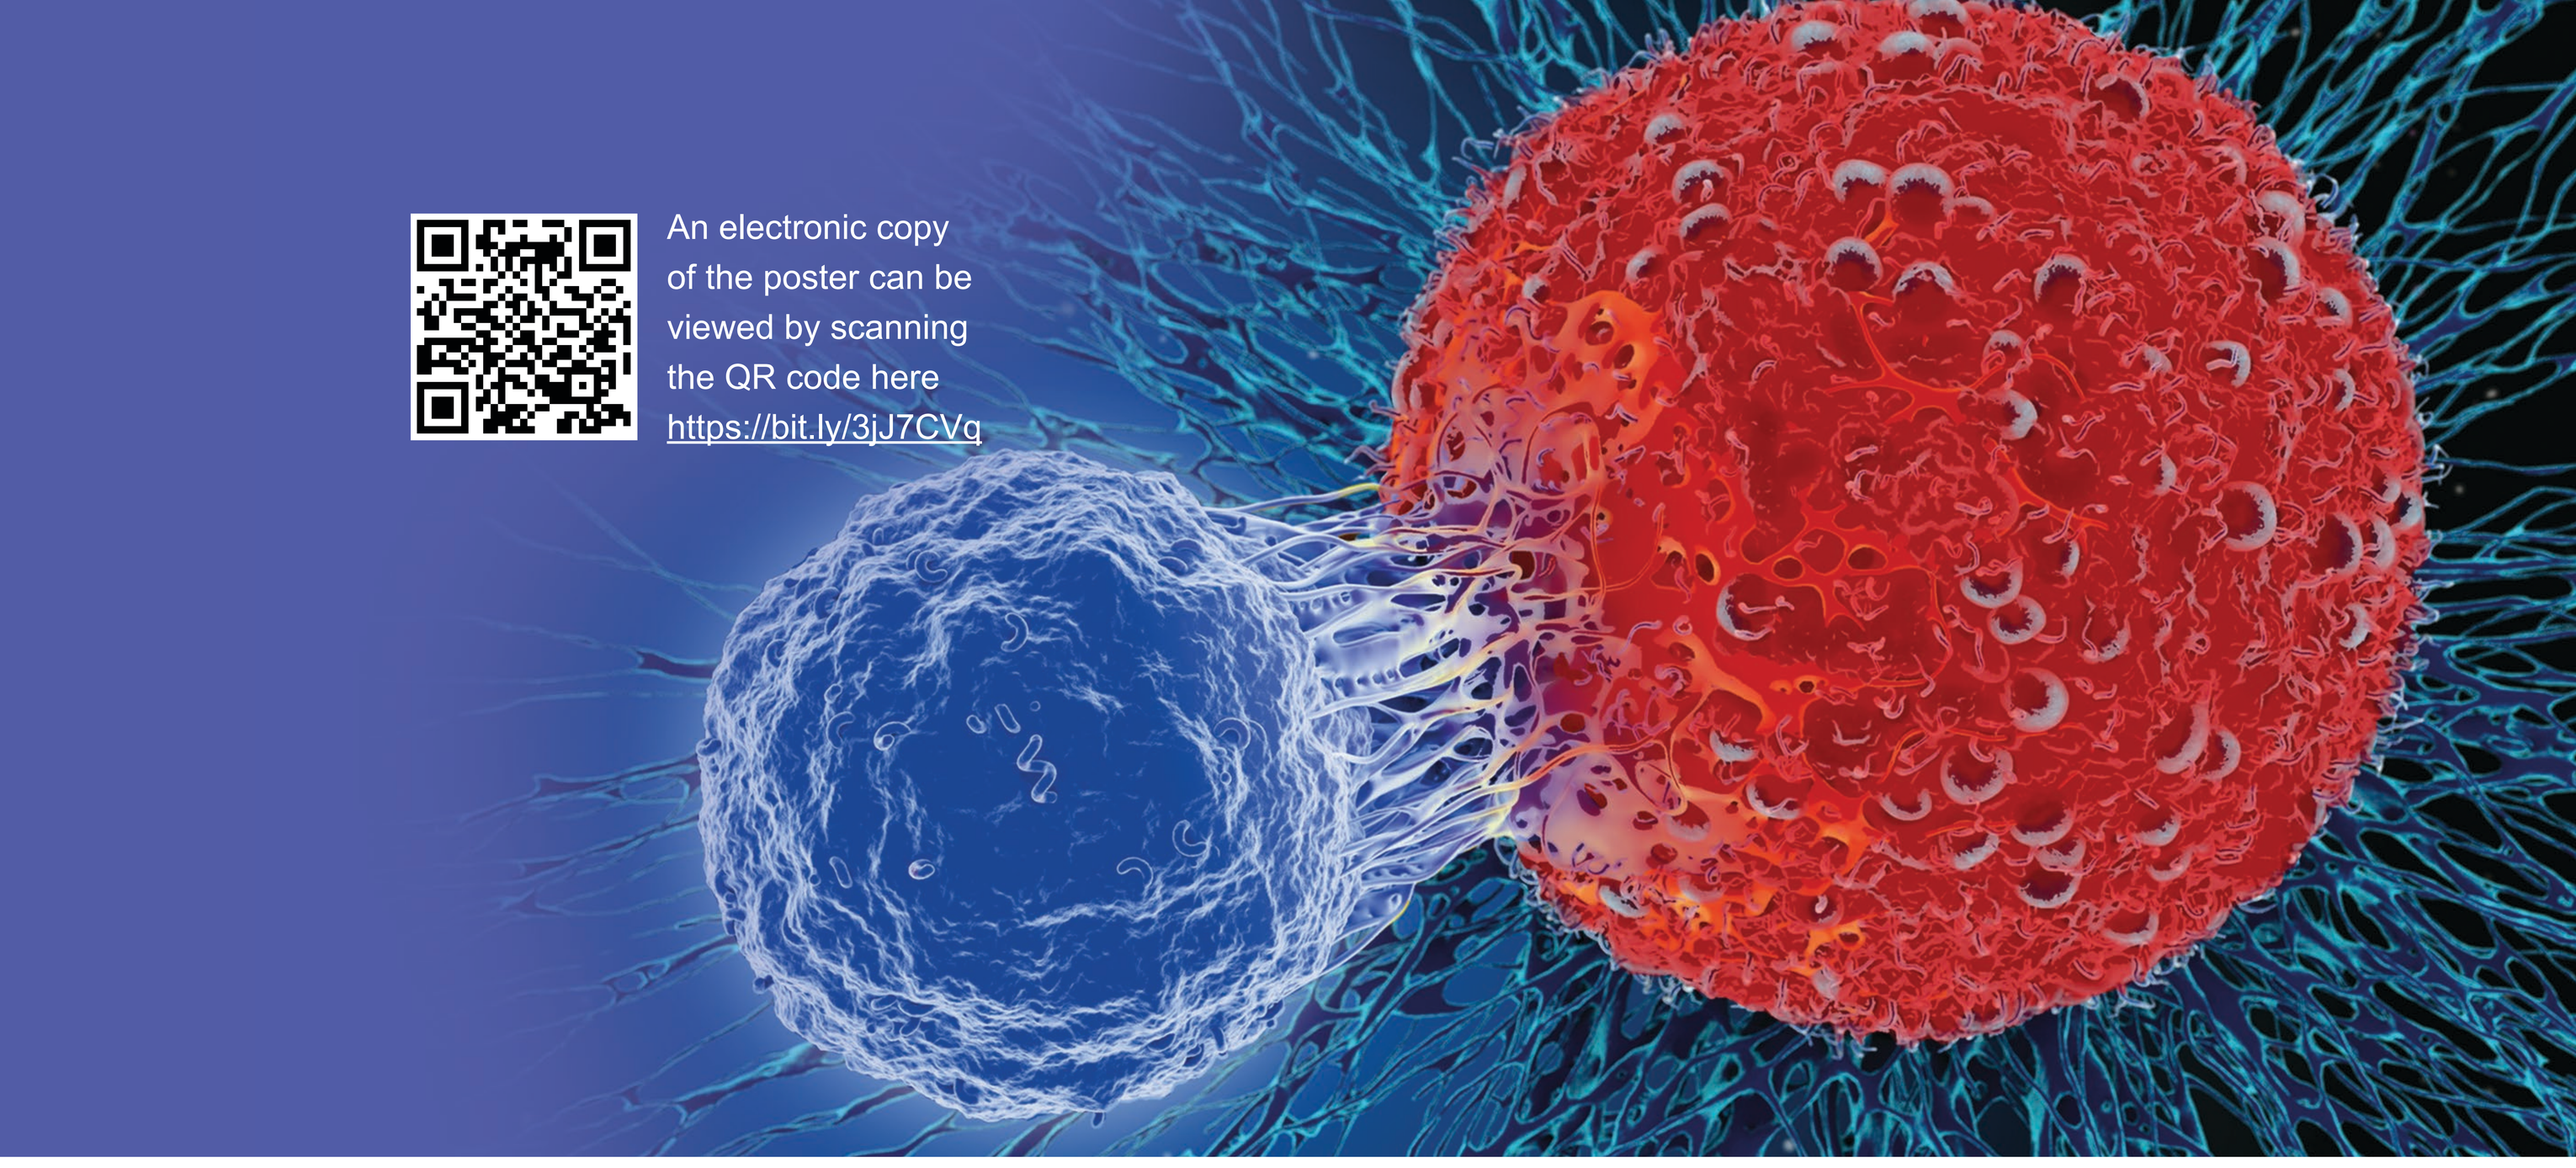

Introduction

- T-cell attributes for adoptive cell therapy for patients with advanced cancer can be optimized during *ex vivo* expansion culture.
- Inhibition of AKT signaling, driven by T-cell receptor (TCR) activation and cytokine stimulation during *ex vivo* expansion process, has been hypothesized to uncouple TCR activation-induced proliferation and terminal differentiation programs, leading to generation of less differentiated T-cells with increased functional potential.[1,2]
- Inhibition of AKT signaling during T-cell expansion has been previously shown to generate T-cells with enhanced anti-tumor effector function and stem cell-like properties, resulting in increased persistence *in vivo* during adaptive T-cell transfer.[3,4]
- Both TCR and cytokine growth factor signals used for *ex vivo* T-cell expansion promote robust activation of AKT (protein kinase B) signaling, which drives T-cell activation, proliferation, and terminal differentiation.

Mechanism of Action

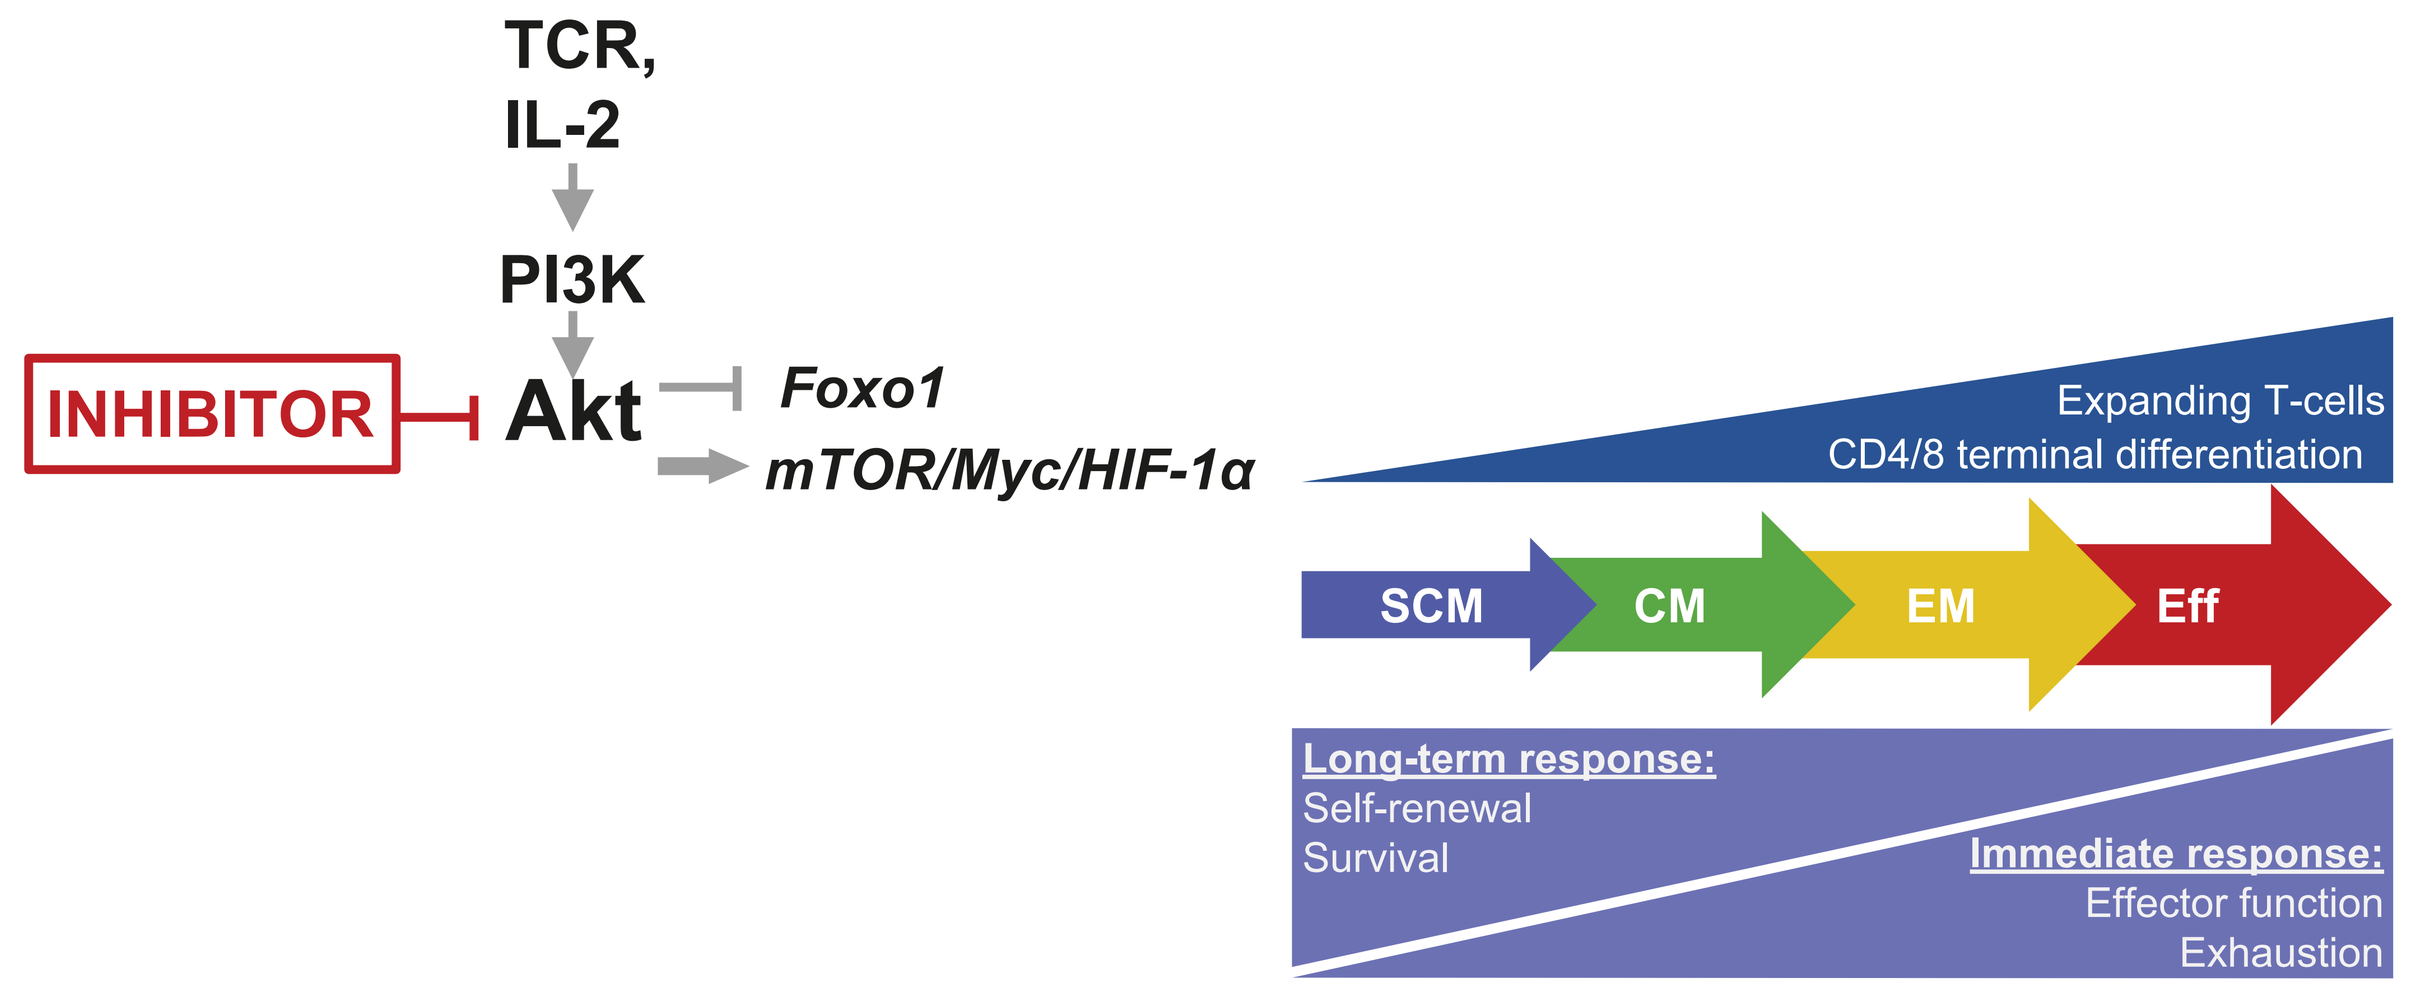
TCR,
IL-2
PI3K
INHIBITOR
Akt
Foxo1
mTOR/Myc/HIF-1α
Expanding T-cells
CD4/8 terminal differentiation
SCM
CM
EM
Eff
Long-term response:
Self-renewal
Survival
Immediate response:
Effector function
Exhaustion

CM, central memory; Eff, effector; EM, effector memory; SCM, stem cell memory.

Objectives

- To determine if the addition of an AKT inhibitor (AKTi) during manufacture of Adaptimmune's proprietary specific peptide enhanced affinity receptor (SPEAR) T-cells leads to the generation of less differentiated SPEAR T-cells that may have increased functional potential.

Figure 1. AKT Inhibition During SPEAR T-Cell Manufacture Provides More Robust Expansion and Memory Phenotype of the Final Product

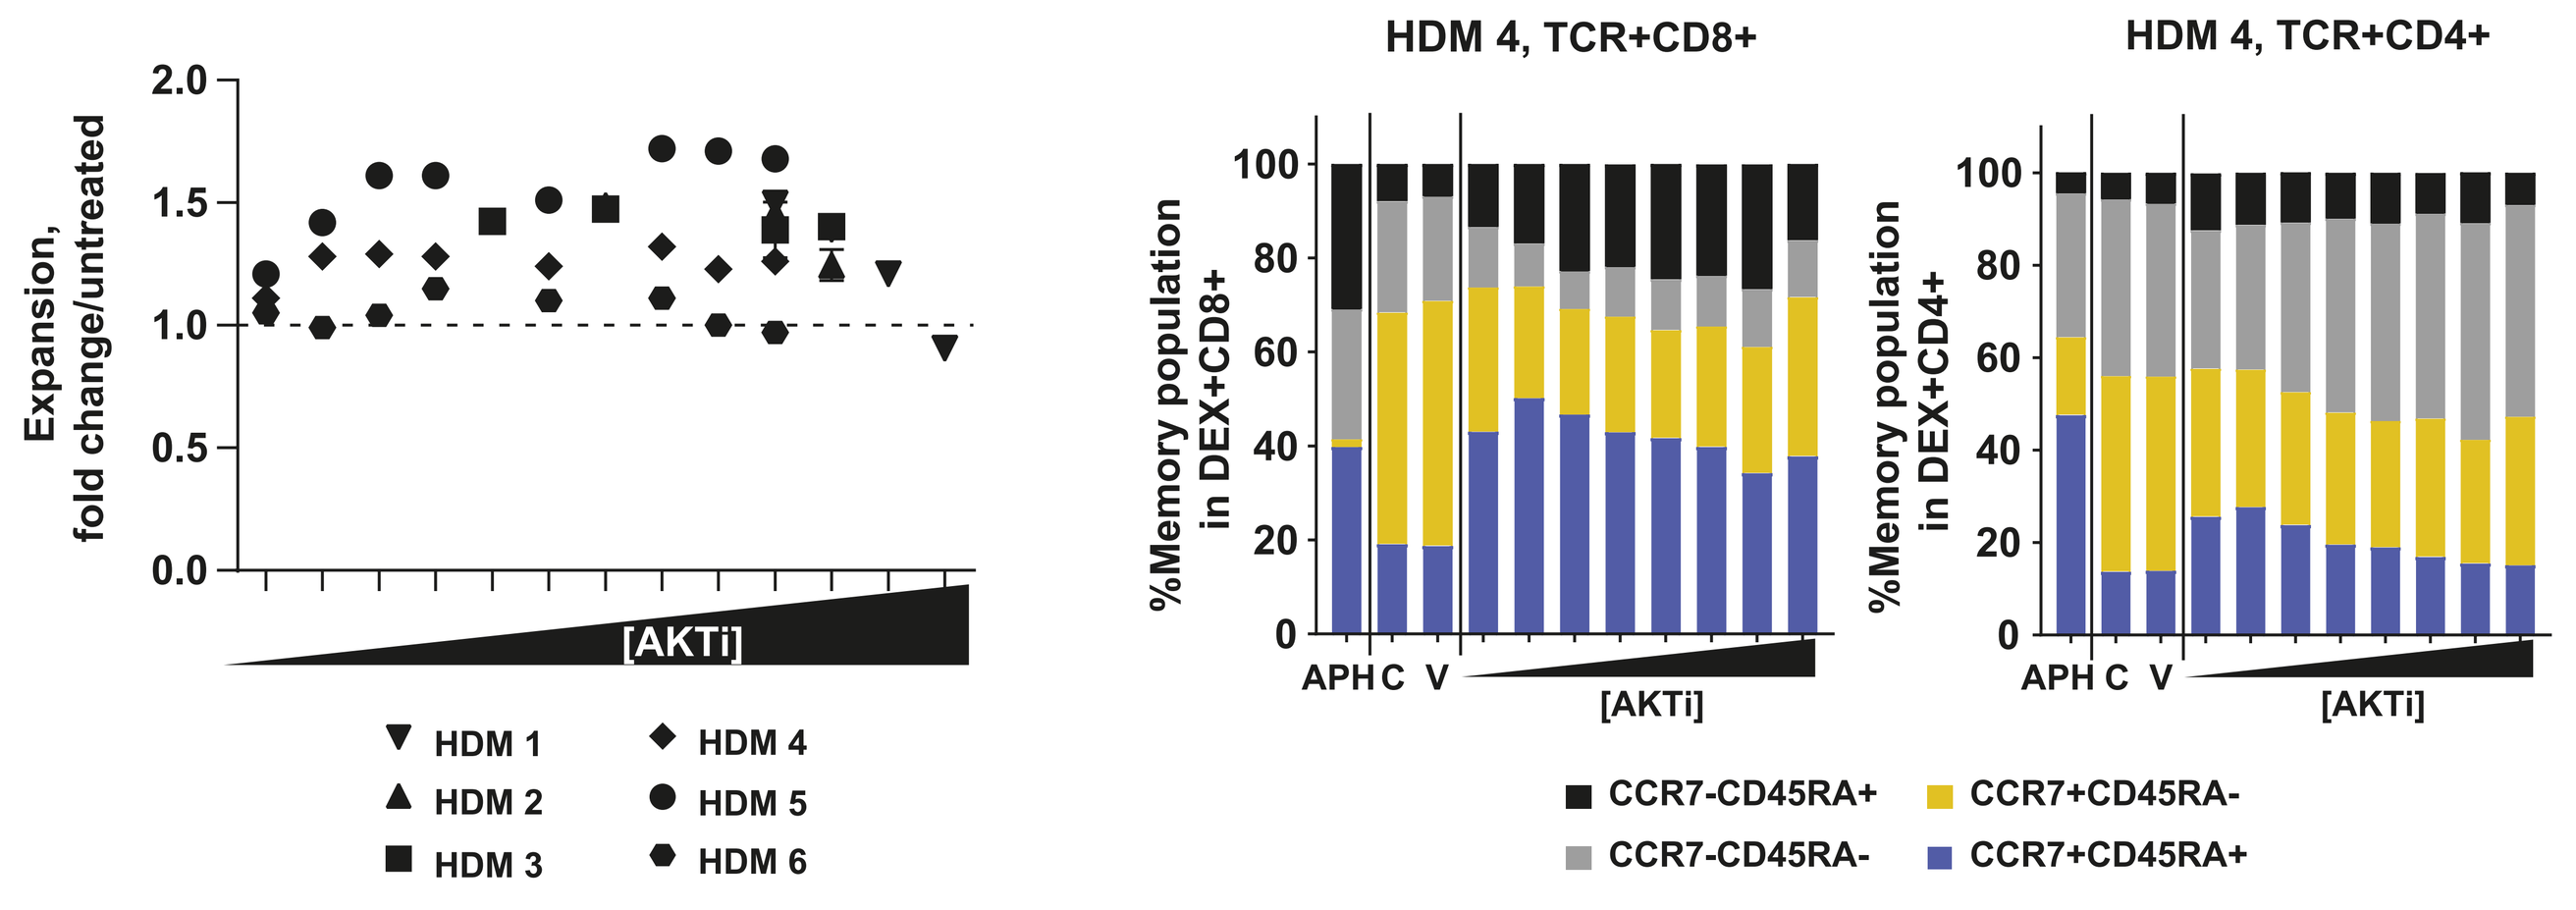
Expansion,
fold change/untreated
[AKTi]
HDM 1
HDM 2
HDM 3
HDM 4
HDM 5
HDM 6
HDM 4, TCR+CD8+
HDM 4, TCR+CD4+
%Memory population
in DEX+CD8+
%Memory population
in DEX+CD4+
APH C V
CCR7-CD45RA+
CCR7-CD45RA-
CCR7+CD45RA-
CCR7+CD45RA+

Six healthy donor leukapheresis material (HDM) were expanded in the presence of increasing concentrations of AKT inhibitor side-by-side with untreated control. Overall expansion in the presence of AKTi was plotted as fold difference from untreated control (dotted line) for each donor.
CCR7/CD45RA expression was analysed by flow cytometry on gated CD8+ and CD4+ T-cells in starting apheresis material or transduced expanded CD8+ T-cells (TCR+CD8+) and CD4+ T-cells (TCR+CD4+). Representative data are shown for HDM4 donor.
AKTi, AKT inhibitor; APH, apheresis material; C, untreated control; HDM, healthy donor material; TCR, T-cell receptor, V, vehicle-alone control.

Figure 2. No Negative Impact on ADP-A2M4 and ADP-A2M4CD8 SPEAR T-Cell Final Product Attributes Following AKT Inhibition During *Ex Vivo* Expansion

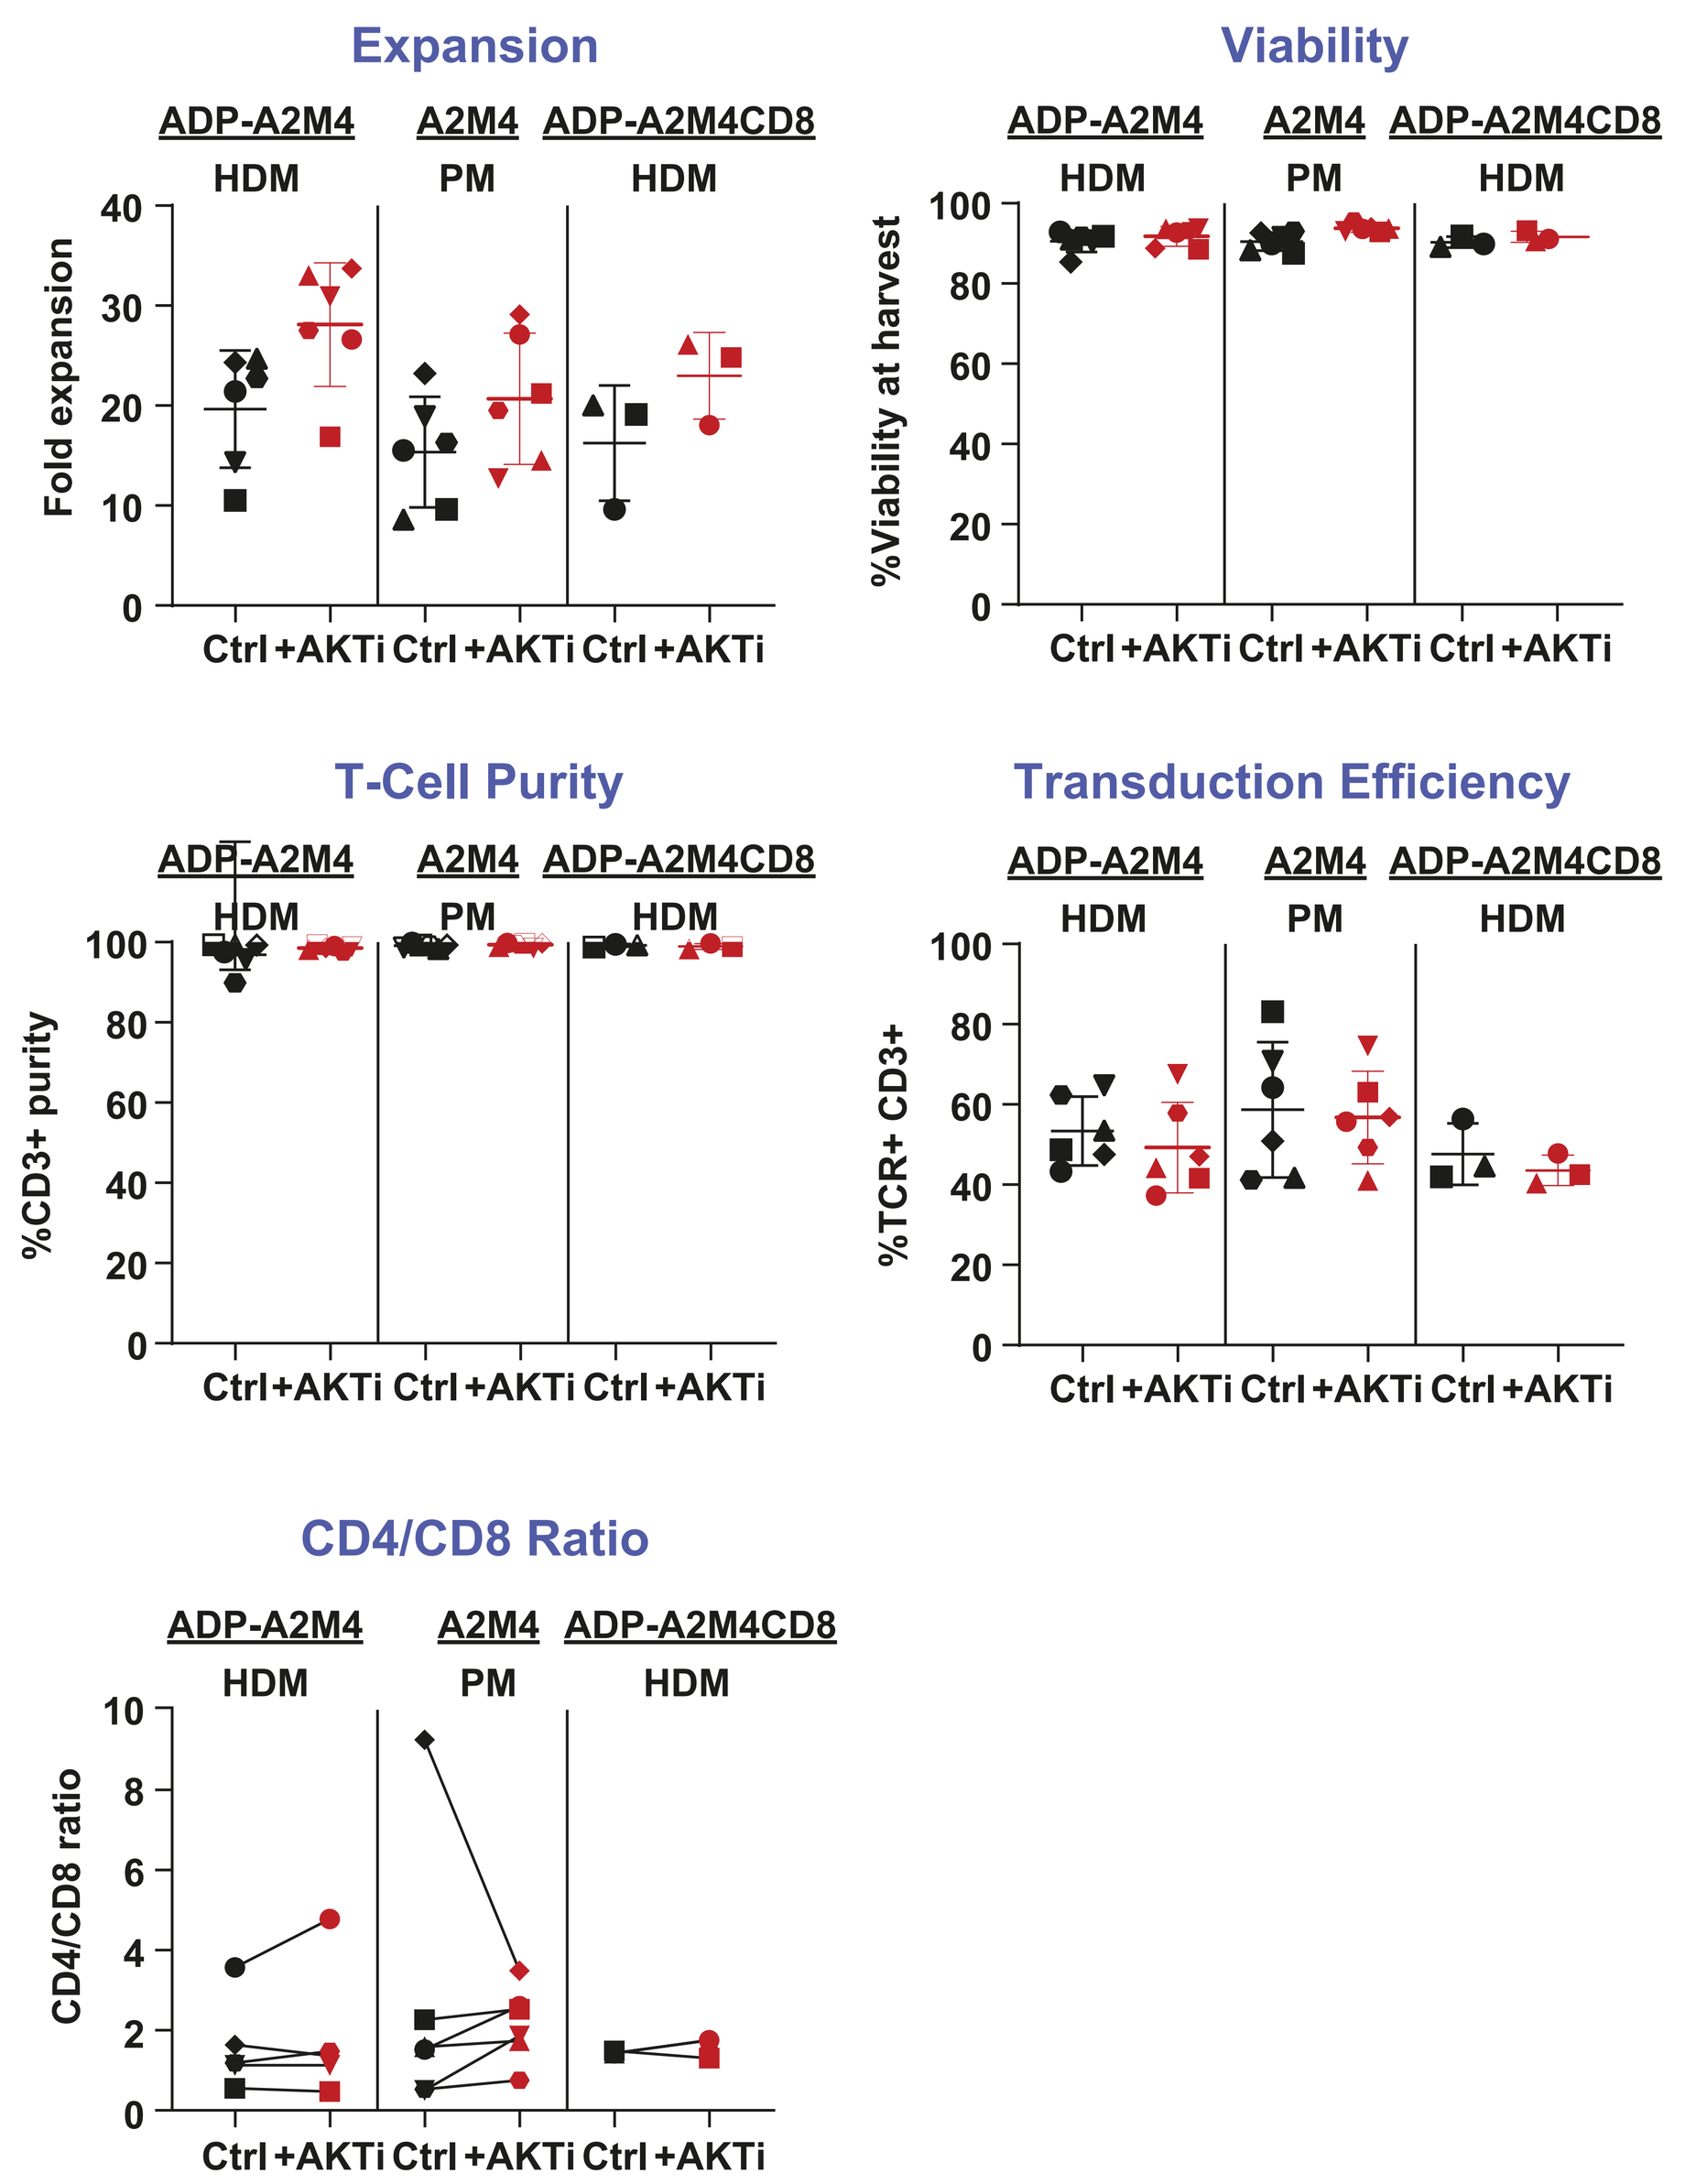
Expansion
Viability
T-Cell Purity
Transduction Efficiency
CD4/CD8 Ratio
ADP-A2M4
HDM
A2M4
PM
ADP-A2M4CD8
HDM
Fold expansion
%Viability at harvest
%CD3+ purity
%TCR+ CD3+
CD4/CD8 ratio
Ctrl +AKTi Ctrl +AKTi Ctrl +AKTi

All donors (healthy donor material, patient material) were expanded with or without AKT inhibitor (red) side-by-side. Expansion was determined as fold difference in T-cell numbers between seeding and harvest. Viability was determined by trypan-blue exclusion method. T-cell purity was determined by flow cytometry as %CD3+ T-cell population frequency in harvest product. CD4/CD8 ratio and transduction efficiency of CD3+ T-cells were measured by flow cytometry immediately after harvest.
AKTi, AKT inhibitor; Ctrl, untreated control; HDM, healthy donor material; PM, patient material.
Data show values for individual donors +SD between donors.

Figure 3. Inhibition of AKT During SPEAR T-Cell Expansion Maintains Less Differentiated Status of CD8+ Population in the Final Product (ADP-A2M4 and ADP-A2M4CD8)

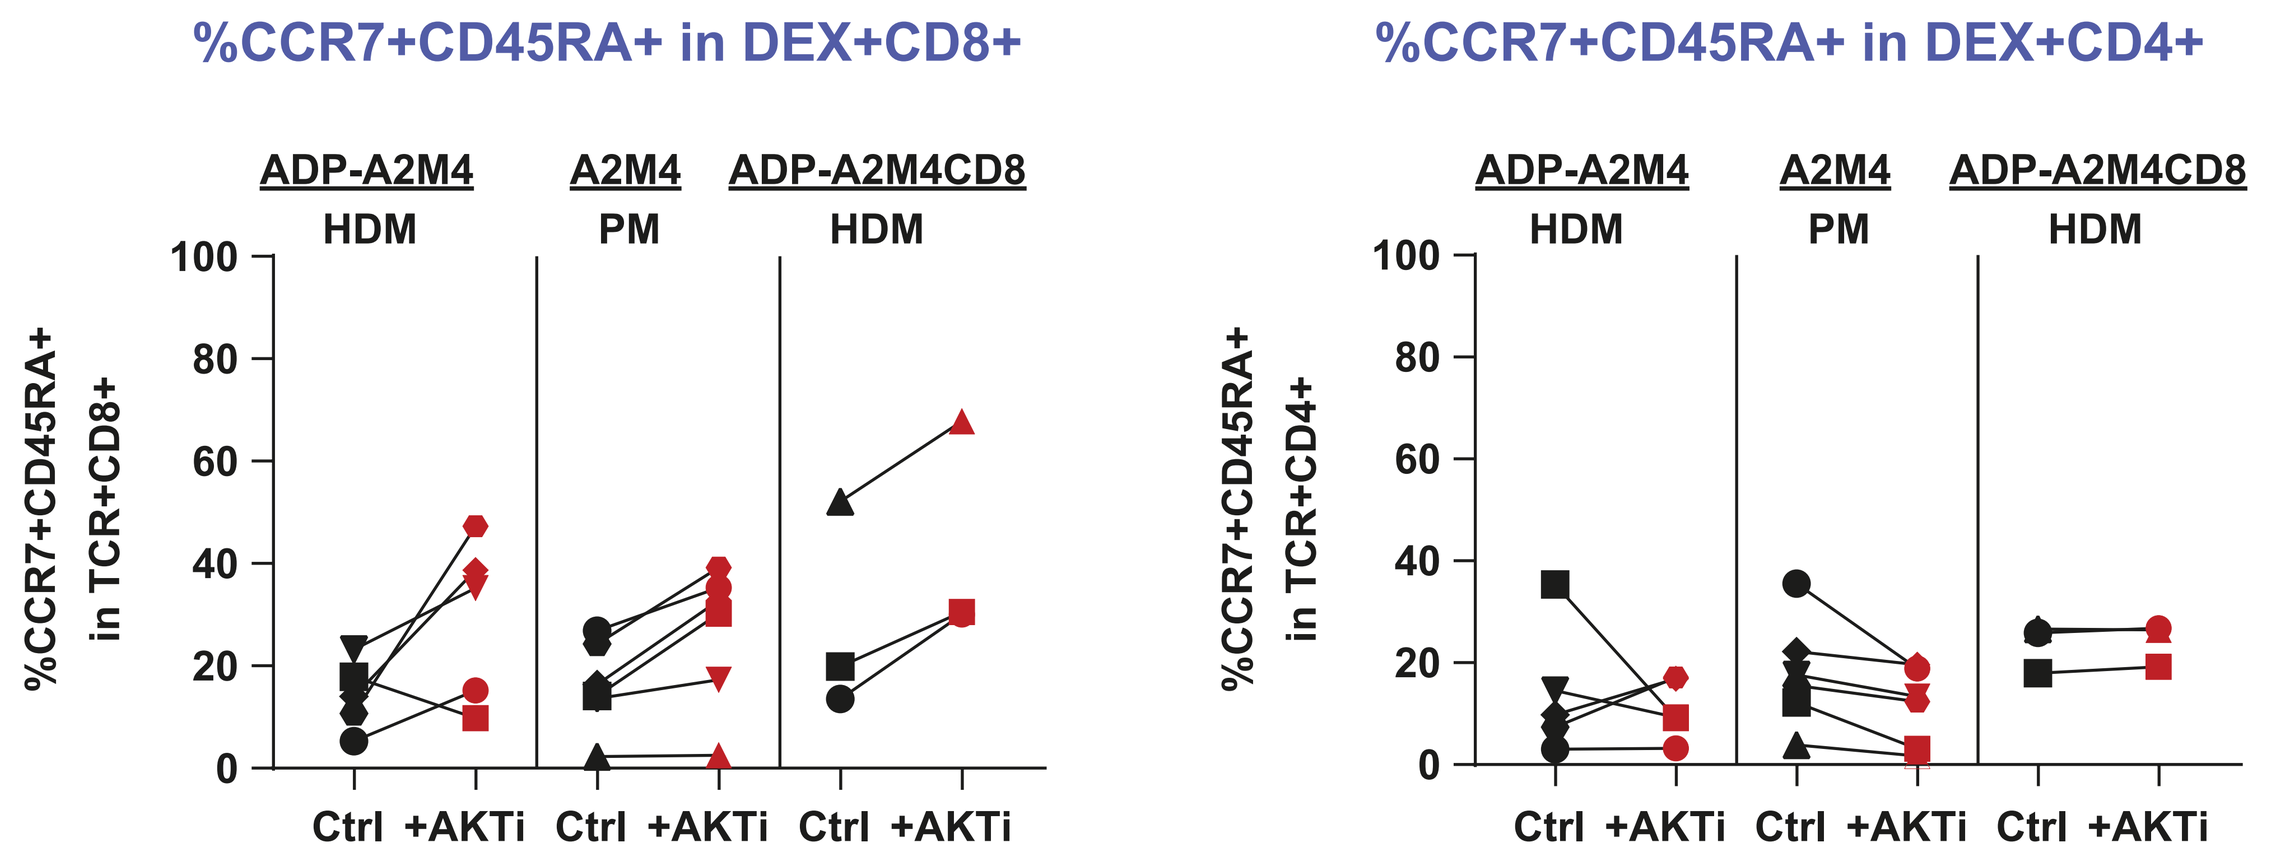
%CCR7+CD45RA+ in DEX+CD8+
%CCR7+CD45RA+ in DEX+CD4+
ADP-A2M4
HDM
A2M4
PM
ADP-A2M4CD8
HDM
%CCR7+CD45RA+
in TCR+CD8+
%CCR7+CD45RA+
in TCR+CD4+
Ctrl +AKTi Ctrl +AKTi Ctrl +AKTi

CCR+CD45RA+ cells are the more naïve / less differentiated population of T-cells.
Differentiation/memory phenotype of expanded SPEAR T-cells was determined by flow cytometry based on CCR7/CD45RA expression. CCR7+CD45RA+ population was consistently increased in expanded transduced CD8+ T-cells (TCR+CD8+) but not in expanded transduced CD4+ T-cells (TCR+CD4+) in the presence of AKT inhibitor (red) compared to untreated control.
AKTi, AKT inhibitor; Ctrl, untreated control; HDM, healthy donor material; PM, patient material, TCR, T-cell receptor.

Figure 4. AKT inhibition During SPEAR T-cell Expansion Generates T-cells with Increased Capability for Antigen-Stimulated IFN-γ and IL-2 Secretion

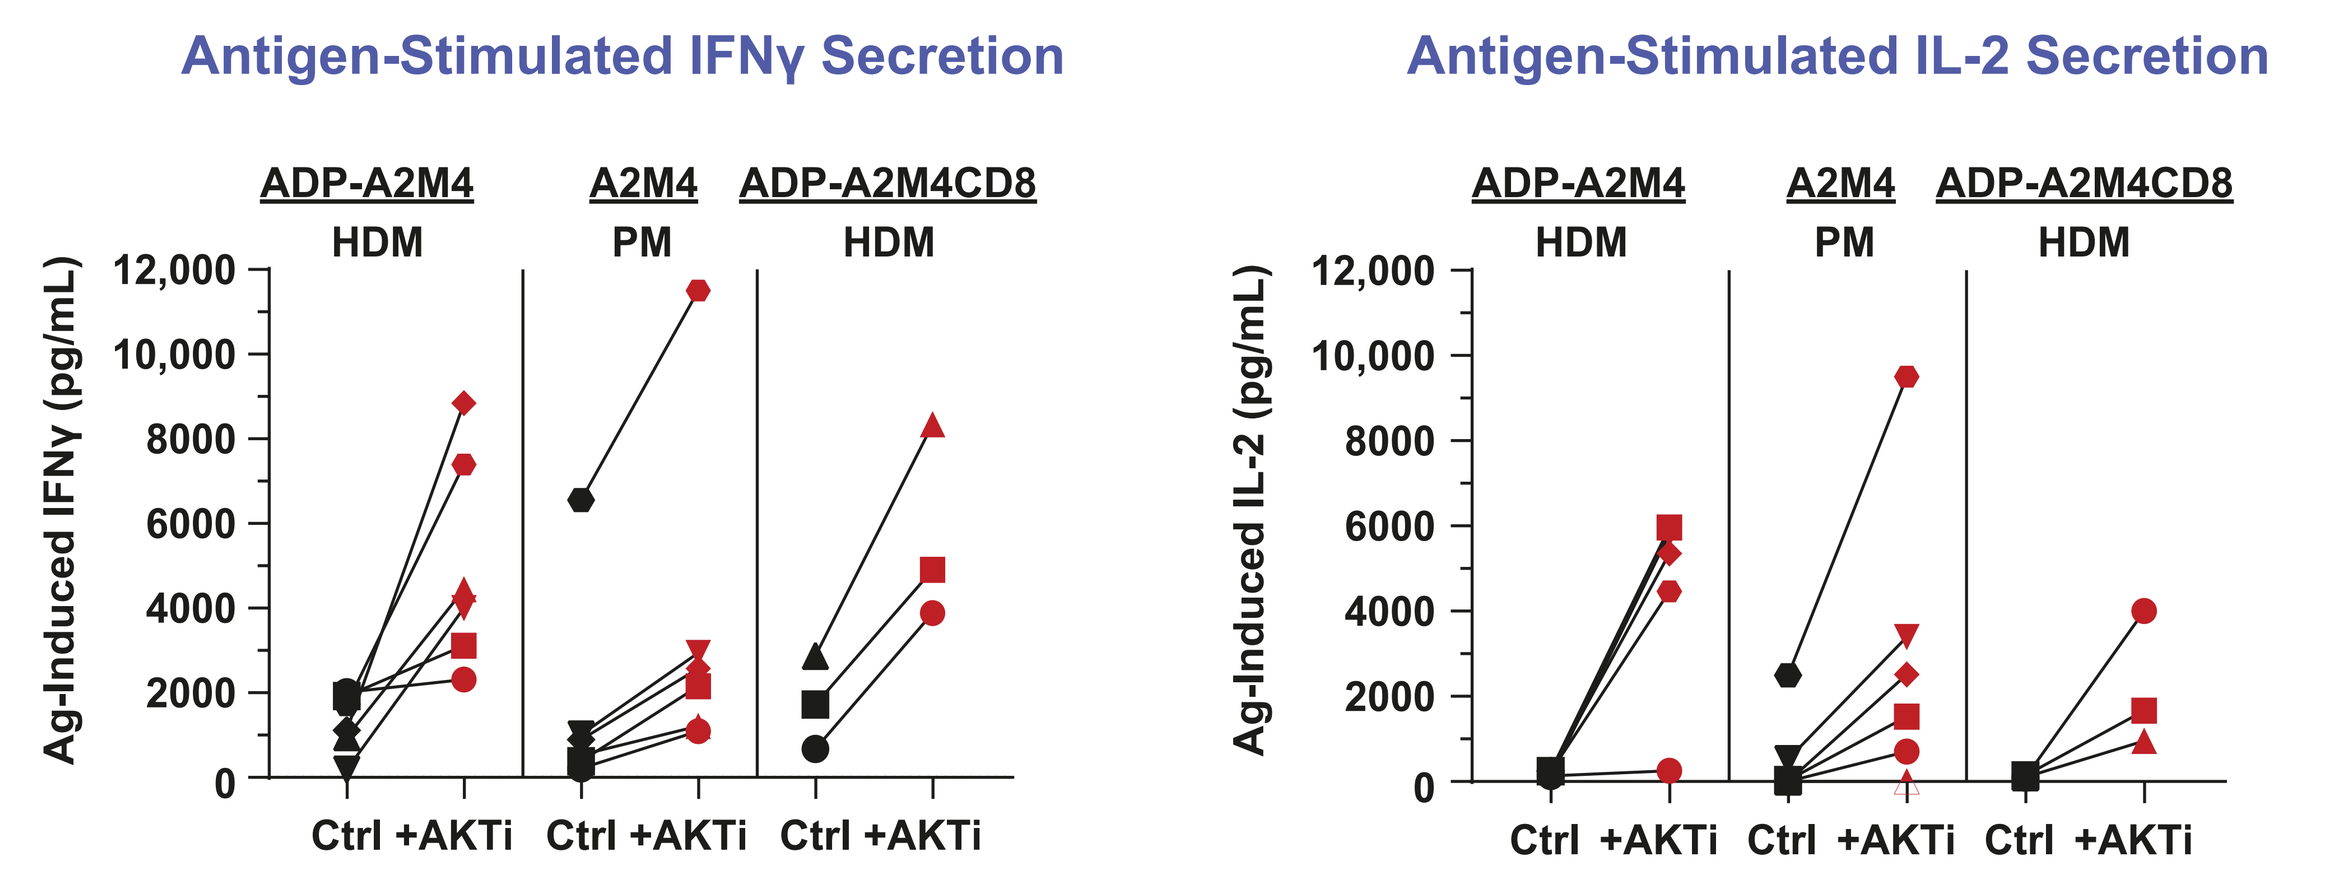
Antigen-Stimulated IFNγ Secretion
Antigen-Stimulated IL-2 Secretion
ADP-A2M4
HDM
A2M4
PM
ADP-A2M4CD8
HDM
Ag-Induced IFNγ (pg/mL)
Ag-Induced IL-2 (pg/mL)
Ctrl +AKTi Ctrl +AKTi Ctrl +AKTi

ADP-A2M4 and ADP-A2M4CD8 T-cells expanded with AKTi (red) or without (Ctrl) AKT inhibitor were stimulated with A375 (MAGE-A4+HLA-A*02+) target cell line for 48 hours. Secretion of IFNγ and IL-2 levels was measured by enzyme-linked immunosorbent assay.
Non-specific cytokine secretion was not detected in corresponding non-transduced T-cell controls (data not shown).
AKTi, AKT inhibitor; Ctrl, untreated control; HDM, healthy donor material; PM, patient material.

Figure 5. AKT Inhibition During SPEAR T-Cell Expansion Generates T-Cells with Increased Antigen-Specific Killing Effector Function

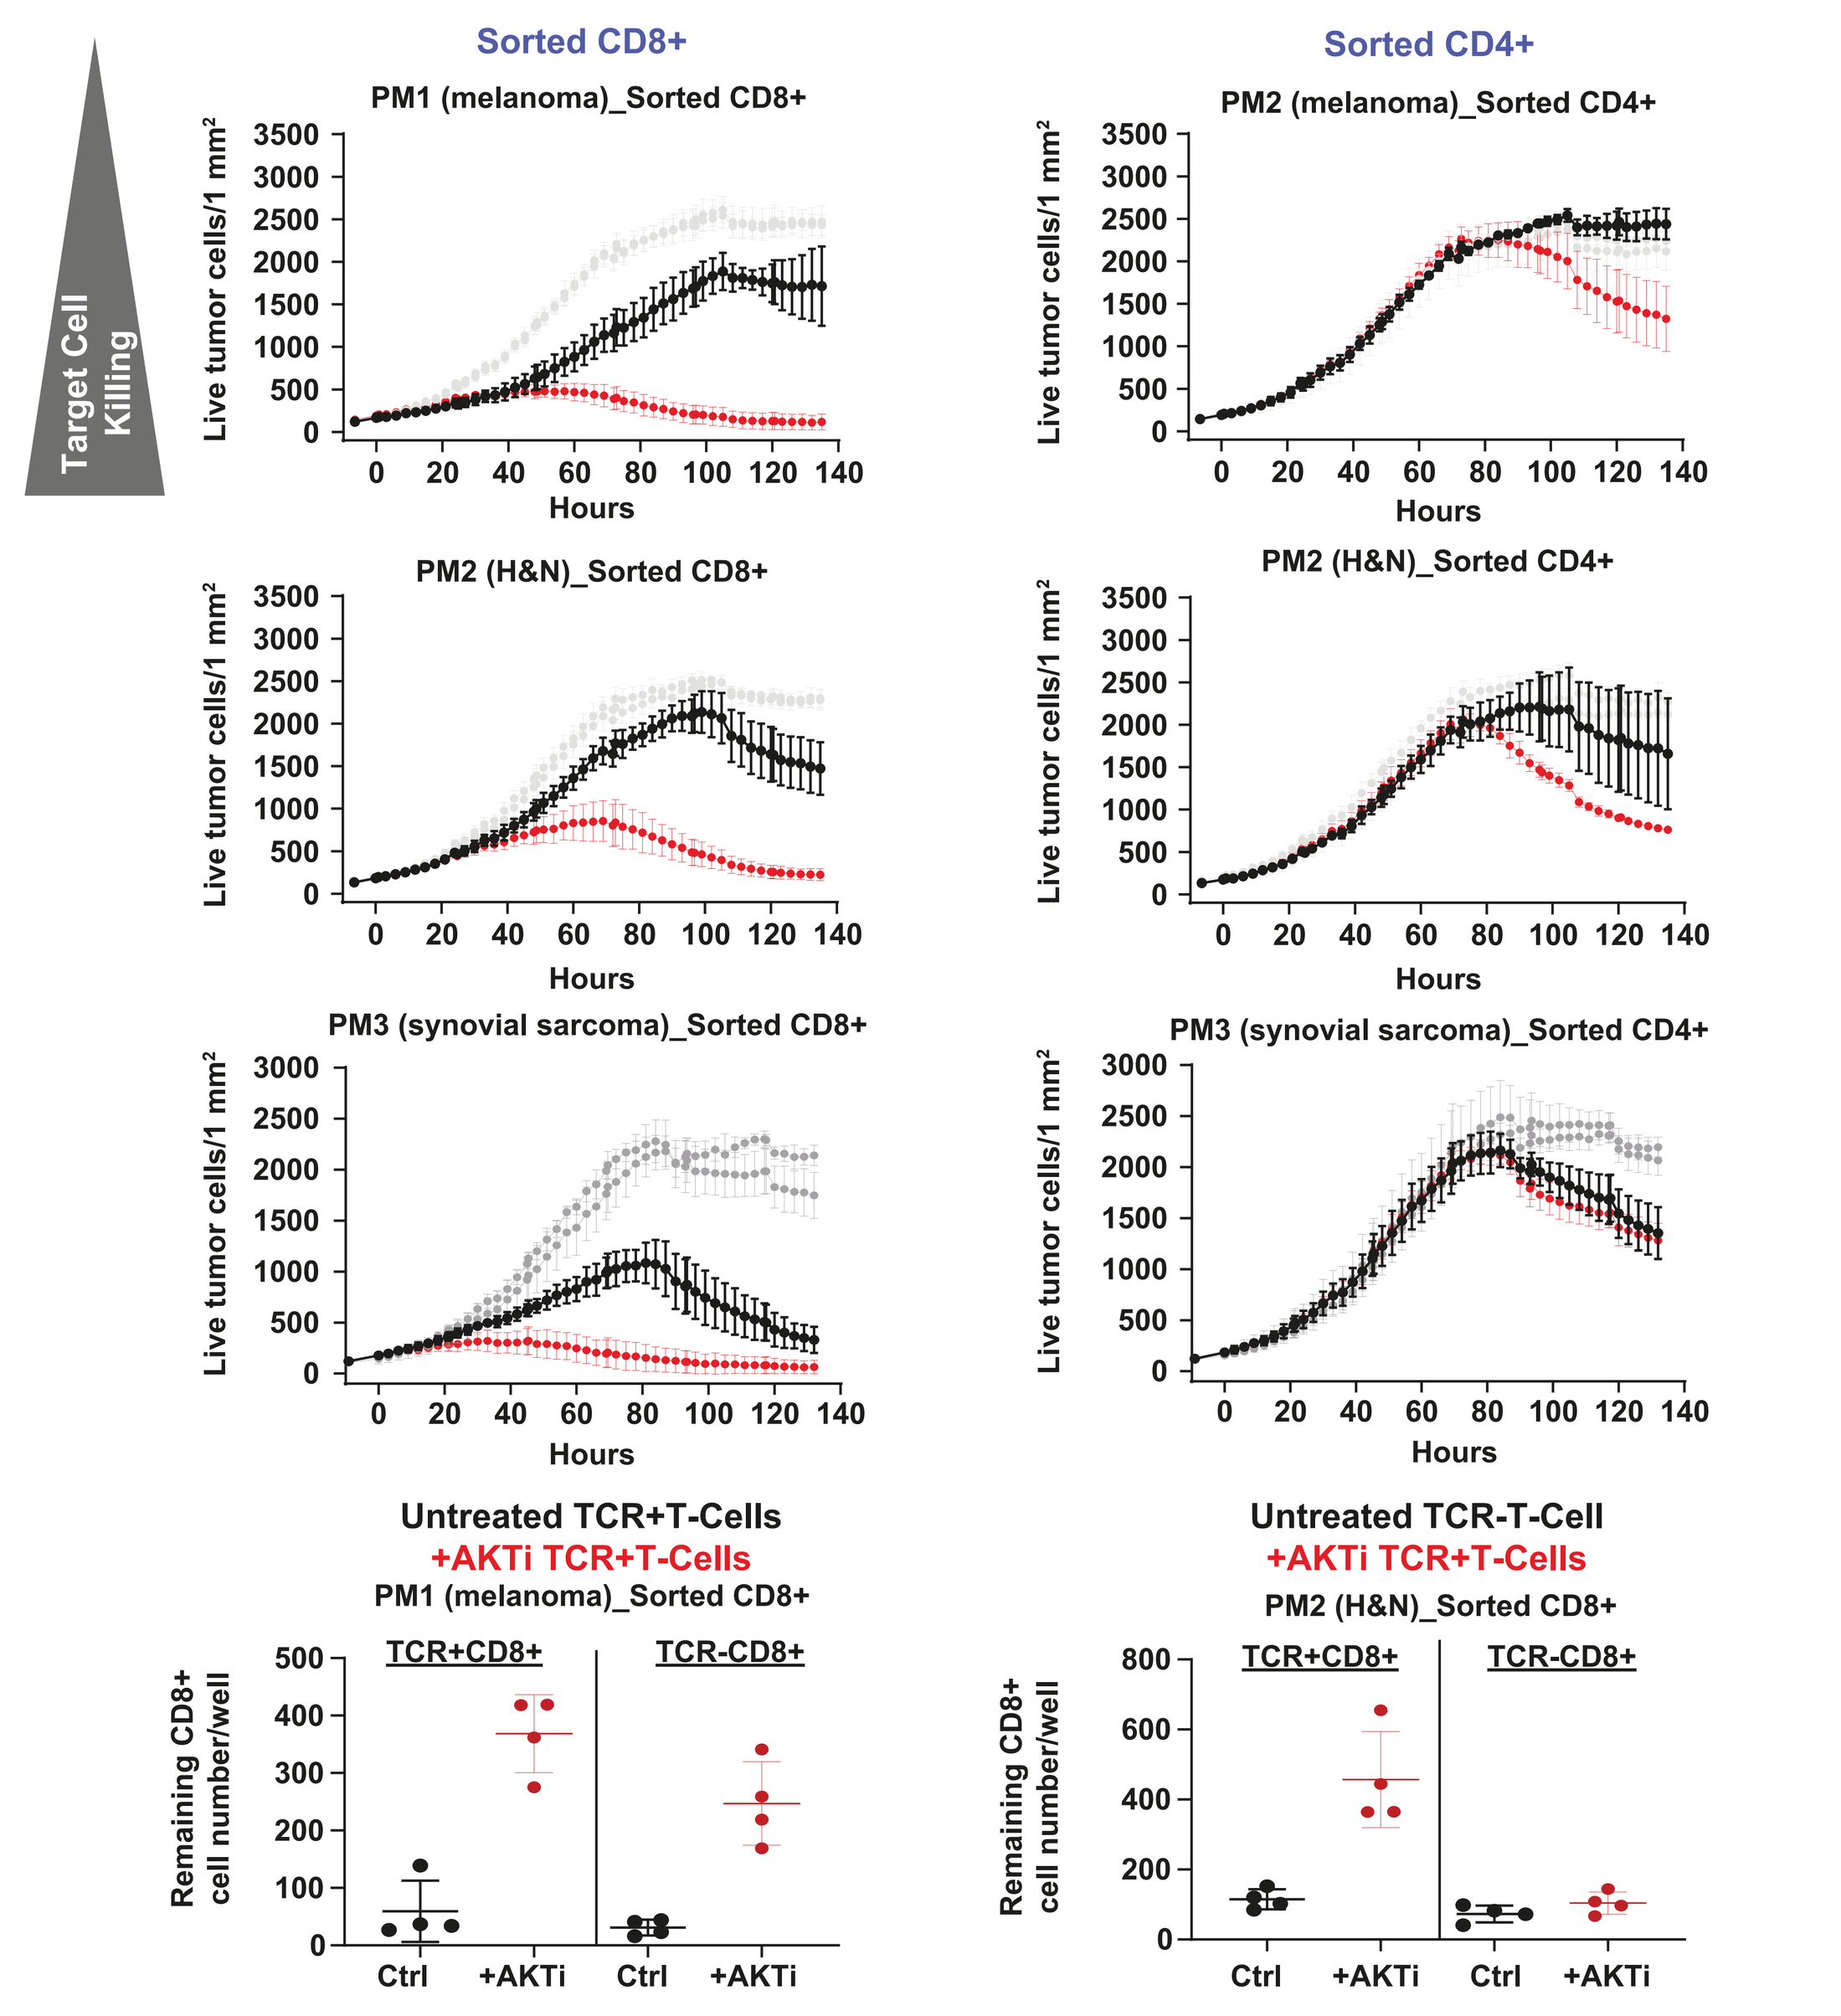
Target Cell
Killing
Sorted CD8+
Sorted CD4+
PM1 (melanoma)_Sorted CD8+
PM2 (melanoma)_Sorted CD4+
PM2 (H&N)_Sorted CD8+
PM2 (H&N)_Sorted CD4+
PM3 (synovial sarcoma)_Sorted CD8+
PM3 (synovial sarcoma)_Sorted CD4+
Live tumor cells/1 mm²
Hours
Untreated TCR+T-Cells
+AKTi TCR+T-Cells
PM1 (melanoma)_Sorted CD8+
Untreated TCR-T-Cell
+AKTi TCR+T-Cells
PM2 (H&N)_Sorted CD8+
TCR+CD8+
TCR-CD8+
Remaining CD8+
cell number/well
Ctrl +AKTi Ctrl +AKTi

ADP-A2M4 T-cells were sorted for transduced CD8+ (TCR+CD8) and CD4+ (TCR+CD4+) populations, and for non-transduced CD8+ (TCR-CD8+) and CD4+ (TCR-CD4+) populations. Sorted T-cells were seeded in the presence of GFP-expressing A375 (MAGE-A4+HLA-A*02+) target cell line. Antigen-specific target cell killing by ADP-A2M4 T-cells was monitored in 384-well format IncuCyte assay by quantifying GFP+ target cell numbers over 7 days. Data show mean values of 4 replicate wells per time point +/-SD.
At the end of the assay, the numbers of CD8+ and CD4+ cells (transduced TCR+, and non-transduced TCR-) remaining in each well were counted by flow cytometry using counting beads. Each data point shows cell count value for a replicate well; error bars show SD between replicate wells.
AKTi, AKT inhibitor (red); Ctrl, untreated control; GFP, green fluorescent protein; PM, patient material.

Figure 6. AKT Inhibition During SPEAR T-cell Expansion Generates T-Cells with Increased Proliferative Capacity in Response to Prolonged/Repeated Stimulation with Antigen-Positive Target Cells

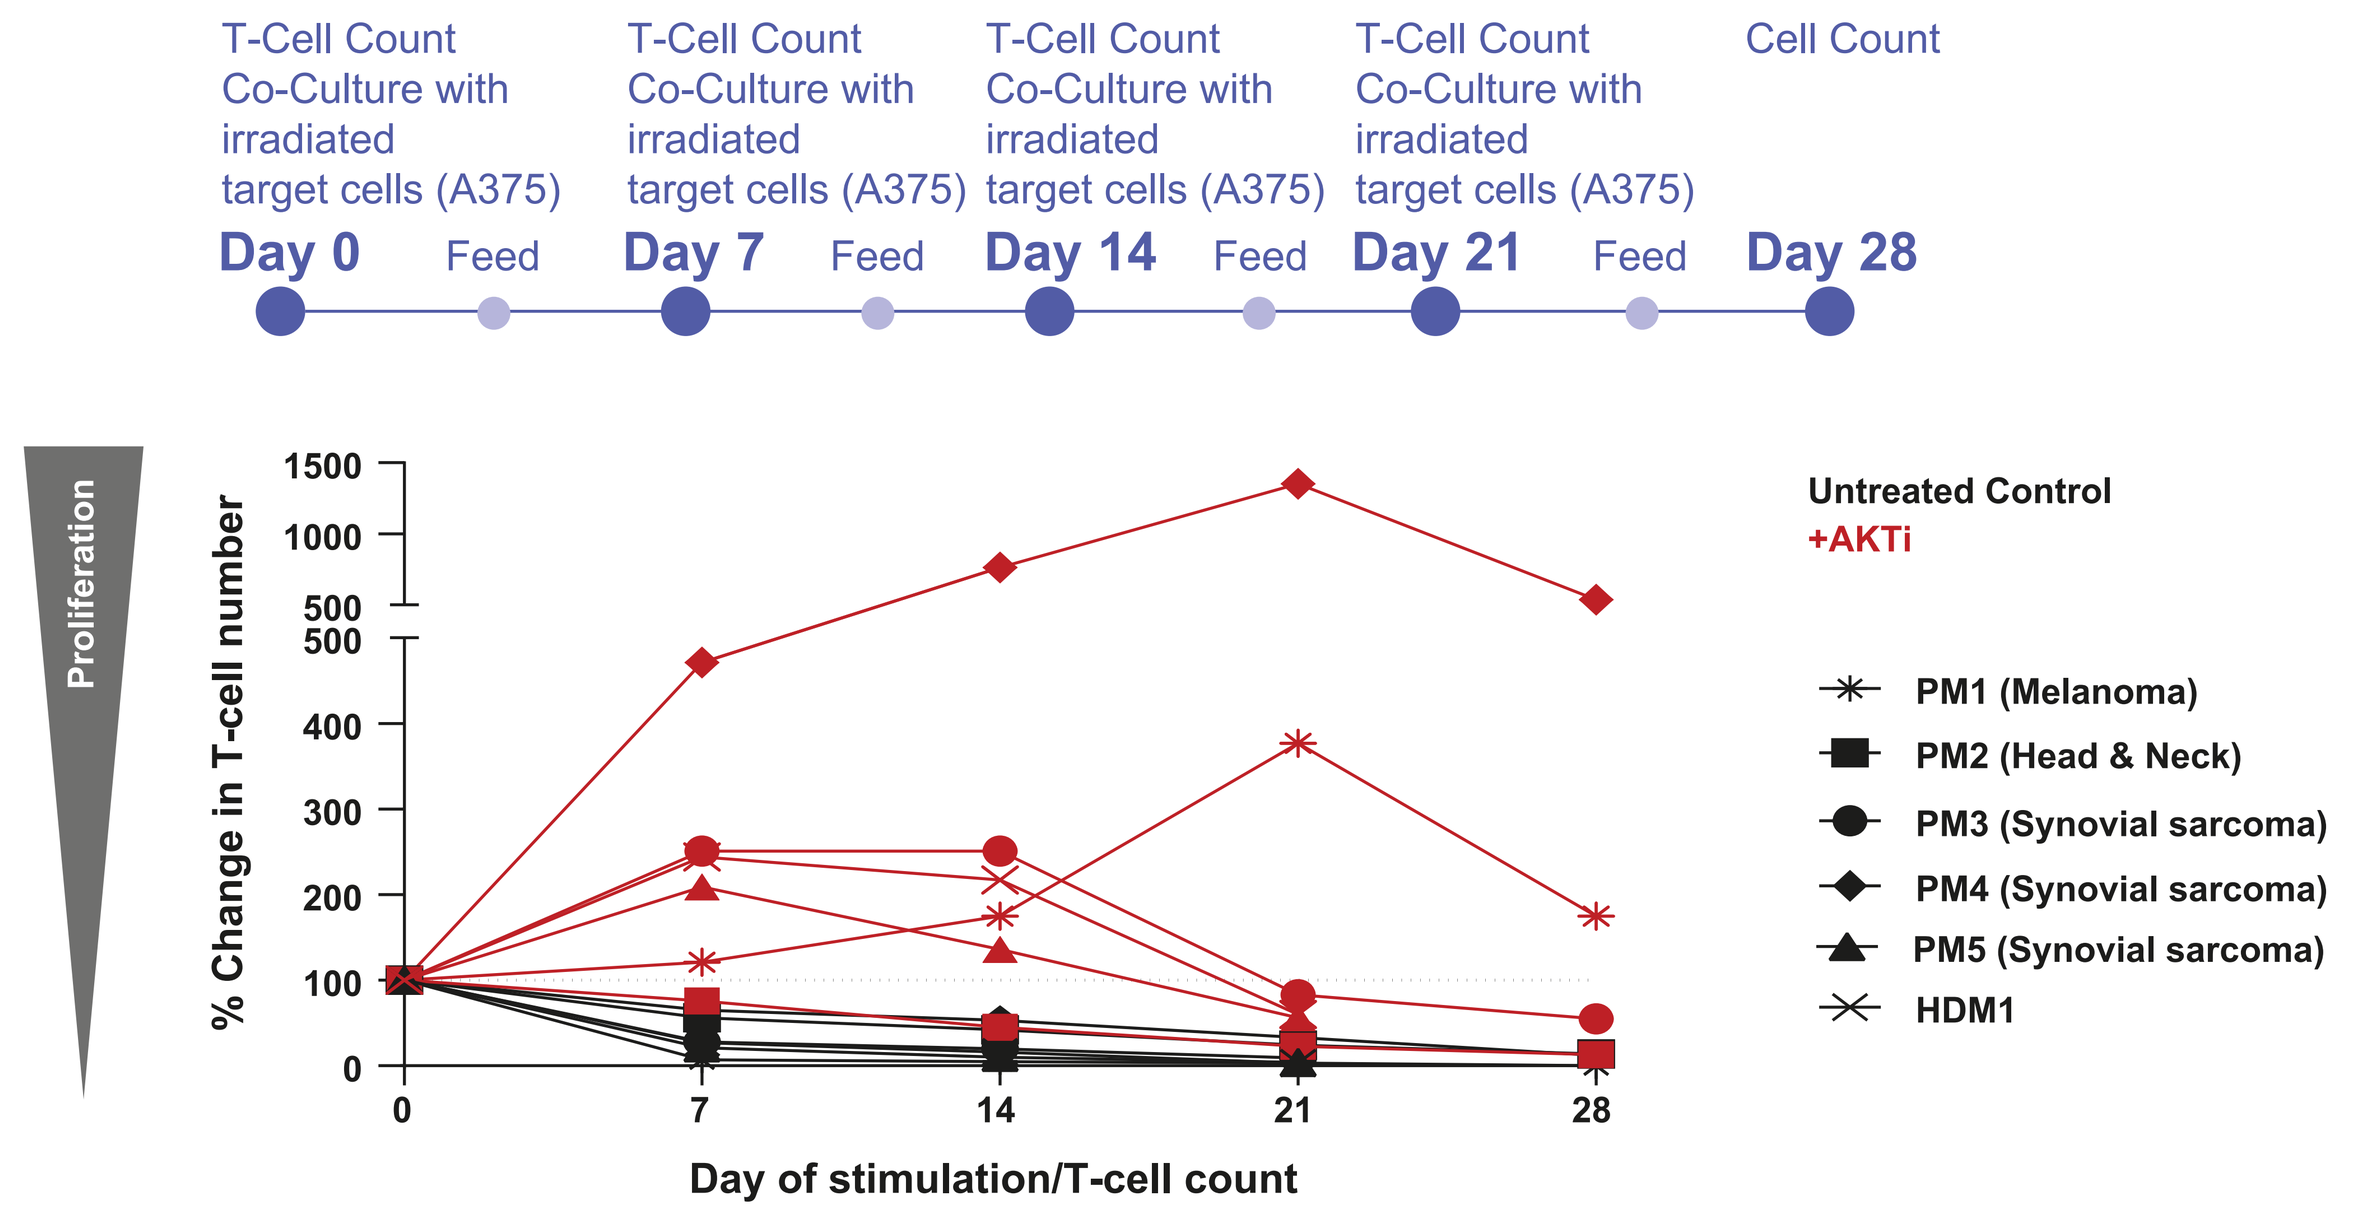
T-Cell Count
Co-Culture with
irradiated
target cells (A375)
Cell Count
Day 0
Feed
Day 7
Day 14
Day 21
Day 28
Proliferation
% Change in T-cell number
Day of stimulation/T-cell count
Untreated Control
+AKTi
PM1 (Melanoma)
PM2 (Head & Neck)
PM3 (Synovial sarcoma)
PM4 (Synovial sarcoma)
PM5 (Synovial sarcoma)
HDM1

ADP-A2M4 T-cells were stimulated with irradiated A375 (MAGE-A4+) target tumor cells for repeated 7-day cycles for up to 28 days. Data show percent change in live T-cell number between weekly harvest and prior seeding.
AKTi, AKT inhibitor; HDM, healthy donor material; PM, patient material.

Conclusions

- AKT inhibition during manufacture of Adaptimmune's proprietary SPEAR T-cells resulted in a more consistent expansion and phenotype of the final product, while having no negative impact on other attributes of both ADP-A2M4 and ADP-A2M4CD8 T-cell product.
- AKT inhibition during manufacturing consistently maintained a higher percentage of CCR7+CD45RA+, less differentiated CD8+ T-cells, resulting in an increased frequency of CCR7+CD45RA+ CD8+ cells in the final ADP-A2M4 and ADP-A2M4CD8 products.
 - Phenotype of expanded CD4+ T-cells was not affected by AKT inhibition during SPEAR T-cell expansion.
- AKT inhibition during SPEAR T-cell expansion consistently generated T-cells with increased magnitude and duration of antigen-dependent responses *in vitro*:
 - Increased antigen-induced secretion of IFNγ and IL-2
 - Additional profiling of 25 cytokines of antigen-stimulated ADP-A2M4 and ADP-A2M4CD8 T-cells did not identify other cytokines or chemokines with strong or consistent increase in SPEAR T-cells expanded in the presence of AKTi, compared to untreated control (data not shown)
 - Increased target cell killing by expanded transduced CD8+ T-cells
 - Antigen-specific proliferation during prolonged/repeated antigen stimulation
- Therefore, Adaptimmune may further consider introduction of AKT inhibition into its GMP manufacturing process. Subsequent use of AKT inhibition may lead to further process development, clinical and regulatory updates, and evaluation of the efficacy of resulting products in ongoing clinical trials.

Acknowledgements and Disclosures

We are extremely grateful to the patients, who were previously enrolled in our clinical trials, and healthy donors for their consent for R&D studies. This was a collaborative cross-functional project, and we are grateful for the contributions of the following scientists: Garth Hamilton, Adel Toth, Abigail Kay, Sophie Badie, Josh Griffiths, Kaushik Sarkar, and Anoop Chandran

K Mardilovich is employed by and has stock or other ownership interests in Adaptimmune

Editorial support and formatting assistance were provided by Debra Brocksmith, MB ChB, PhD, of Elevate Scientific Solutions, which was contracted and compensated by Adaptimmune for these services

E-poster: Society for Immunotherapy of Cancer (SITC), Virtual Congress, November 9–14, 2020
Presenter contact information: Katerina Mardilovich, Katerina.Mardilovich@adaptimmune.com

References

1. Utzschneider DT, et al. *Cell Reports.* 2018;22:3454–67.
2. Kim EH & Suresh M. *Front Immunol.* 2013;4:20; doi:10.3389/fimmu.2013.00020.
3. Klebanoff CA, et al. *JCI Insight.* 2017;2:e95103.
4. van der Waart AB, et al. *Blood.* 2014;124:3490–500.

SPEAR T-cell mechanism of action video can be viewed by scanning the QR code here https://youtu.be/zdI8IGXoQd0