Identification of Response Stratification Factors from Pooled Efficacy Analyses of Afamitresgene Autoleucel (“Afami-cel” [Formerly ADP-A2M4]) in Metastatic Synovial Sarcoma and Myxoid/Round Cell Liposarcoma Phase 1 and Phase 2 Trials

Sandra P. D'Angelo,[1] Steven Attia,[2] Jean-Yves Blay,[3] Sandra J. Strauss,[4] Claudia Maria Valverde Morales,[5] Albiruni Ryan Abdul Razak,[6] Erin Van Winkle,[7] Thejo Annareddy,[7] Chandra Sattigari,[7] Evangelo Diamantopoulos,[7] Dennis Williams,[7] Elliot Norry,[7] Swethajit Biswas,[8] Dejka M. Araujo,[9] Brian Andrew Van Tine[10]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Mayo Clinic, Jacksonville, FL, USA; [3]Centre Léon Bérard, Lyon, France; [4]University College London Hospitals, London, UK; [5]Vall d´Hebron University Hospital, Barcelona, Spain; [6]Princess Margaret Cancer Centre, Toronto, ON, Canada; [7]Adaptimmune, Philadelphia, PA, USA; [8]Adaptimmune, Abingdon, Oxfordshire, UK; [9]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [10]Washington University School of Medicine, St. Louis, MO, USA

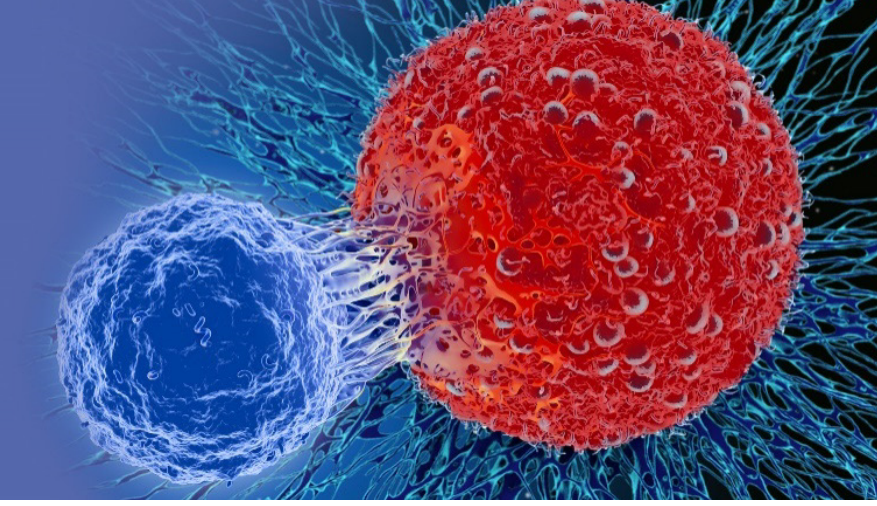

Introduction

- Afamitresgene autoleucel (“afami-cel” [formerly ADP-A2M4]) is an autologous, specific peptide enhanced affinity receptor T-cell therapy engineered to target melanoma-associated antigen A4 (MAGE-A4)-positive solid tumors in human leukocyte antigen (HLA)-A*02-eligible patients.
- There is a significant unmet need for treatment in patients with unresectable or metastatic synovial sarcoma after first-line systemic therapy.
- The pivotal, Phase 2 SPEARHEAD-1 trial (NCT04044768) is evaluating afami-cel in patients with synovial sarcoma or myxoid/round cell liposarcoma (MRCLS) in two cohorts.
 - The trial met its primary endpoint based on Cohort 1 data. As of September 1, 2021, the overall response rate (ORR) in 47 patients per Response Evaluation Criteria in Solid Tumors (RECIST) v1.1 by independent review was 34% with encouraging durability.[1]
- To identify potential stratification factors for response and assess whether response is a proxy for progression-free survival (PFS), we present pooled analyses using data from patients with synovial sarcoma or MRCLS in the prior Phase 1 trial (NCT03132922) of afami-cel and Cohort 1 of the Phase 2 SPEARHEAD-1 trial.

Methods

- The prior Phase 1 dose-escalation and expansion trial of afami-cel evaluated patients who were HLA-A*02 eligible with advanced cancers that expressed MAGE-A4 (NCT03132922):
 - Urinary bladder cancer, melanoma, head and neck cancer, ovarian cancer, non-small cell lung cancer, esophageal cancer, gastric cancer, synovial sarcoma, MRCLS, and esophagogastric junction cancer.
- SPEARHEAD-1 is a two-cohort, single-arm, Phase 2 trial in HLA-A*02-eligible patients with MAGE-A4-positive unresectable or metastatic synovial sarcoma or MRCLS (NCT04044768).
- In the pooled dataset from both trials, tumors were considered MAGE-A4 positive if ≥30% cancer cells had 2+ or 3+ staining intensity using an immunohistochemistry clinical trial assay (CTA), except for 13 patients with synovial sarcoma in the Phase 1 trial who were deemed MAGE-A4 positive with a CTA cut-off of 10%; MAGE-A4 tumor expression was evaluated as a histoscore (H-score[a] range: 0–300).
- Prior to afami-cel infusion (**Figure 1**), patients received a lymphodepletion (LD) chemotherapy regimen of fludarabine 30 mg/m^2 x4 days and cyclophosphamide 600 mg/m^2 x3 days, except for 4 patients with synovial sarcoma (Phase 1 trial) who received fludarabine 30 mg/m^2 x4 days and cyclophosphamide 1800 mg/m^2 x2 days.
- The pooled analyses reported here evaluated ORR per RECIST v1.1 by investigator review in patients with synovial sarcoma and MRCLS, stratified by 9 clinical subgroups. Safety was also investigated.
 - Data cut off was September 1, 2020 for Phase 1 trial and October 11, 2021 for SPEARHEAD-1
 - All analyses utilized the modified intention-to-treat (mITT) population, defined as all patients who received infusion of afami-cel.

Figure 1. Phase 1 and Phase 2 Trial Design

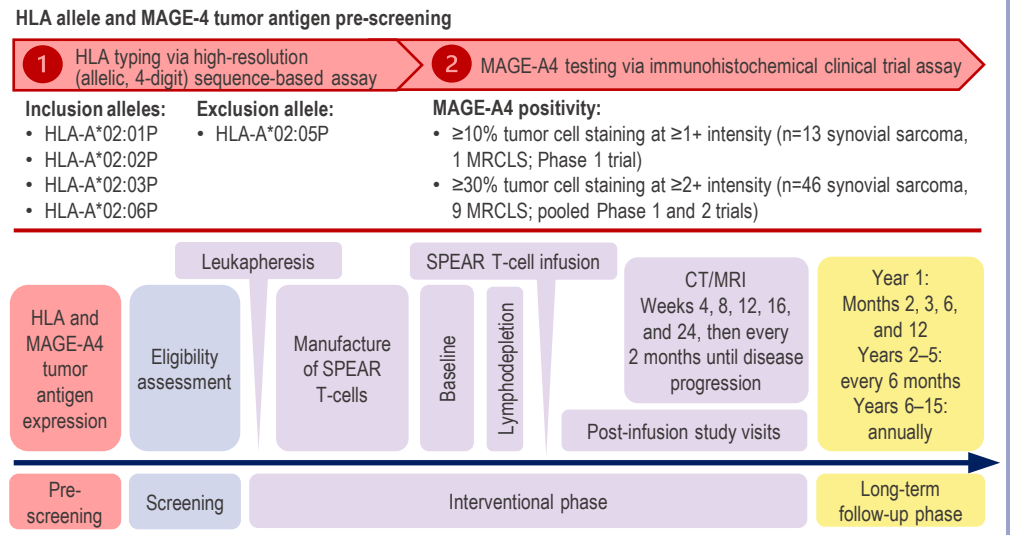
HLA allele and MAGE-4 tumor antigen pre-screening
1 HLA typing via high-resolution (allelic, 4-digit) sequence-based assay
2 MAGE-A4 testing via immunohistochemical clinical trial assay
Inclusion alleles:
• HLA-A*02:01P
• HLA-A*02:02P
• HLA-A*02:03P
• HLA-A*02:06P
Exclusion allele:
• HLA-A*02:05P
MAGE-A4 positivity:
• ≥10% tumor cell staining at ≥1+ intensity (n=13 synovial sarcoma, 1 MRCLS; Phase 1 trial)
• ≥30% tumor cell staining at ≥2+ intensity (n=46 synovial sarcoma, 9 MRCLS; pooled Phase 1 and 2 trials)
Leukapheresis
SPEAR T-cell infusion
HLA and MAGE-A4 tumor antigen expression
Eligibility assessment
Manufacture of SPEAR T-cells
Baseline
Lymphodepletion
CT/MRI Weeks 4, 8, 12, 16, and 24, then every 2 months until disease progression
Post-infusion study visits
Year 1: Months 2, 3, 6, and 12 Years 2–5: every 6 months Years 6–15: annually
Pre-screening
Screening
Interventional phase
Long-term follow-up phase

CT, computed tomography; HLA, human leukocyte antigen; MAGE-A4, melanoma-associated antigen A4; MRI, magnetic resonance; SPEAR, specific peptide enhanced affinity receptor.

Demographics and Baseline Characteristics

- In the pooled ITT population, 79 patients (synovial sarcoma, n=65; MRCLS, n=14) were enrolled and underwent leukapheresis; 70 patients (synovial sarcoma, n=60; MRCLS, n=10) received LD chemotherapy.
- In the pooled mITT population, 69 patients (synovial sarcoma, n=59; MRCLS, n=10) received afami-cel (Phase 1, n=18; Phase 2, n=51).
- All patients expressed at least one eligible HLA-A*02 allele.
- Median MAGE-A4 tumor H-score was higher in synovial sarcoma than in MRCLS (**Table 1**).
- Patients were heavily pretreated; median number of prior lines of therapy was 2, and most patients did not require bridging therapy.

Table 1. Pooled Baseline Characteristics of Patients who Received Afami-cel in the Prior Phase 1 Trial and Cohort 1 of the SPEARHEAD-1 Trial

Characteristic	Overall , N=69
Sex, n (%)	
Male	39 (56.5)
Female	30 (43.5)
Age, years, median (min, max)	42 (19, 76)
H-score, median (min, max)	229.67 (60.3, 300.0)
Synovial sarcoma (n=59)	256.5 (60.3, 300.0)
MRCLS (n=10)	179.5 (112.1, 229.7)
ECOG at baseline, n (%)	
0	38 (55.1)
1	30 (43.5)
2[a]	1 (1.4)
Bridging therapy, yes, n (%)	28 (40.6)
Transduced cells, min, max	2.12×10^9, 9.99×10^9
Prior lines of systemic therapy, median (min, max)	2 (1, 12)

[a]After data cut-off (SPEARHEAD-1), patient was confirmed as ECOG 1. ECOG, Eastern Cooperative Oncology Group; MRCLS, myxoid/round cell liposarcoma.

Efficacy

- Pooled investigator-assessed ORR was 36.2% overall (**Figure 2A**, **Table 2**).
 - In synovial sarcoma, ORR was 40.7% (2 complete responses and 22 partial responses including 2 patients who received the higher cyclophosphamide dose).
 - In MRCLS, ORR was 10.0% (1 partial response).
- Median duration of response was 52.0 weeks (**Figure 2B, Table 2**).
- Median PFS overall was 18.3 weeks (20.4 weeks in synovial sarcoma).
- In responders vs non-responders, median PFS was 58.3 vs 12.0 weeks (**Figure 3**; log-rank P<0.0001).
 - In synovial sarcoma, median PFS in responders was 58.3 vs 11.0 weeks in non-responders.
- Responses were observed across all 9 clinical subgroups (**Table 3**).
- Higher response rates were observed in patients with lower baseline tumor burden (sum of diameters <100 mm vs ≥100 mm), lower number of prior lines of systemic therapy (≤2 vs ≥3), and higher MAGE-A4 tumor H-score (≥200 vs <200; **Table 3**).
 - Similar results were observed among subgroups of patients with synovial sarcoma.

Figure 2. Investigator-Assessed Best Overall Response and Duration of Response per RECIST v1.1

- Pooled investigator-assessed ORR (n=69) was 36.2% (95% CI 24.99, 48.69).
 - Synovial sarcoma (n=59), 40.7% (95% CI 28.07, 54.25).

A

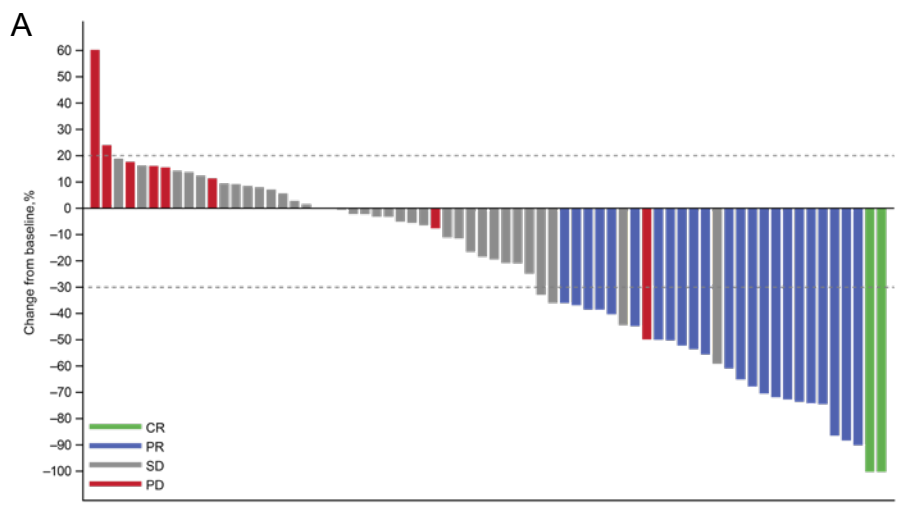
Change from baseline, %
CR
PR
SD
PD

- Median duration of response was 52.0 weeks (95% CI 18.56, N/A).

B

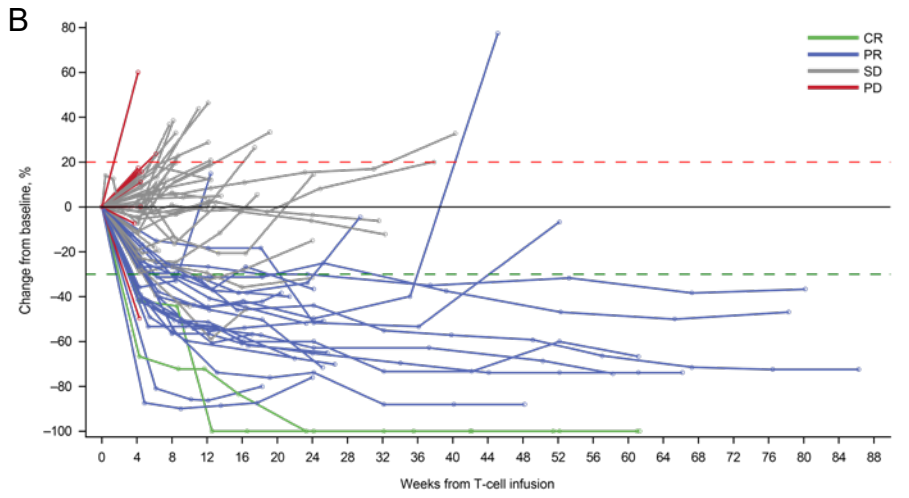
Change from baseline, %
Weeks from T-cell infusion
CR
PR
SD
PD

CR, complete response; ORR, overall response rate; PD, progressive disease; PR, partial response; RECIST, Response Evaluation Criteria in Solid Tumors; SD, stable disease.

Table 2. Responses per RECIST v1.1 by Investigator Review

Response	Overall, N=69
ORR	
n (%) 95% CI[a]	25/69 (36.2) 24.99, 48.69
Complete response	
n (%)	2/69 (2.9)
Partial response	
n (%)	23/69 (33.3)
ORR, synovial sarcoma	
n (%) 95% CI[a]	24/59 (40.7) 28.07, 54.25
Disease control rate[b]	
n (%)	59/69 (85.5)

1 patient was not evaluable per RECIST v1.1. [a]Two-sided 95% CI based on exact Clopper-Pearson (exact Binomial) method. [b]Complete response + partial response + stable disease. CI, confidence interval; ORR, overall response rate; RECIST, Response Evaluation Criteria in Solid Tumors .

Figure 3. Kaplan-Meier Plot of Progression-Free Survival in Responders vs Non-Responders with Synovial Sarcoma or Myxoid/Round Cell Liposarcoma

- Responders (blue), median PFS 58.3 weeks.
- Non-responders (red), median PFS 12.0 weeks.
 - Log-rank test, P<0.0001.

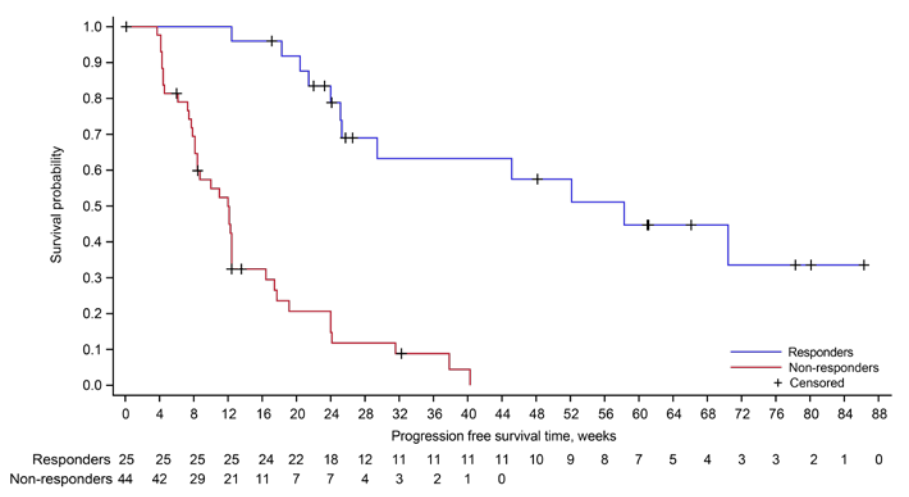
Survival probability
Progression free survival time, weeks
Responders
Non-responders
+ Censored
Responders 25 25 25 25 24 22 18 12 11 11 11 11 10 9 8 7 5 4 3 3 2 1 0
Non-responders 44 42 29 21 11 7 7 4 3 2 1 0

PFS, progression-free survival.

Table 3. Overall Response Rate in Patients with Synovial Sarcoma and Myxoid/Round Cell Liposarcoma Stratified by 9 Clinical Subgroups

Subgroup	Overall response rate	
Baseline sum of diameters	<100 mm, n=34	≥100 mm, n=35
n (%) 95% CI	17/34 (50.0) 32.43, 67.57	8/35 (22.9) 10.42, 40.14
Prior lines of systemic therapy	≤2, n=35	≥3, n=34
n (%) 95% CI	17/35 (48.6) 31.38, 66.01	8/34 (23.5) 10.75, 41.17
H-score	<200, n=26	≥200, n=43
n (%) 95%CI	6/26 (23.1) 8.97, 43.65	19/43 (44.2) 29.08, 60.12
Bridging therapy	Yes, n=28	No, n=41
n (%) 95% CI	8/28 (28.6) 13.22, 48.67	17/41 (41.5) 26.32, 57.89
Transduced cell dose	<7×10^9, n=28	≥ 7×10^9, n=41
n (%) 95% CI	9/28 (32.1) 15.88, 52.35	16/41 (39.0) 24.20, 55.50
Any grade CRS	Yes, n=50	No, n=19
n (%) 95% CI	19/50 (38.0) 24.65, 52.83	6/19 (31.6) 12.58, 56.55
Age	<40 years, n=27	≥ 40 years, n=42
n (%) 95% CI	9/27 (33.3) 16.52, 53.96	16/42 (38.1) 23.57, 54.36
Sex	Male, n=39	Female, n=30
n (%) 95% CI	12/39 (30.8) 17.02, 47.57	13/30 (43.3) 25.46, 62.57
Geographical region	North America, n=55	Europe, n=14
n (%) 95% CI	21/55 (38.2) 25.41, 52.27	4/14 (28.6) 8.39, 58.10

CRS, cytokine release syndrome.

Safety

- Any grade and Grade ≥3 adverse events occurred in all patients.
- Scan QR code (bottom of poster) to view table of treatment-emergent adverse events occurring in ≥10% of patients.
- Treatment-emergent serious adverse events occurring in ≥5% of patients were cytokine release syndrome (CRS; 15.9%), pyrexia (7.2%), and pleural effusion (5.8%).
- Adverse events of special interest (**Table 4**) included CRS, prolonged cytopenia defined as any Grade ≥3 anemia, neutropenia, or thrombocytopenia at Week 4 post T-cell infusion, and immune effector cell-associated neurotoxicity syndrome (ICANS).
- One patient in the Phase 1 trial who received LD chemotherapy cyclophosphamide 1800 mg/m^2 x2 days had Grade 5 treatment-related pancytopenia on Day 55 caused by aplastic anemia, which led to reduction in cyclophosphamide to 600 mg/m^2 x3 days for the remainder of the Phase 1 trial and for SPEARHEAD-1.

Table 4. Summary of Adverse Events of Special Interest

Preferred term	Overall, N=69
Cytokine release syndrome	
Any grade, n (%)	50 (72.5)
Grade ≥3, n (%)	3 (4.3)
Time to onset, days, median (min, max)	2.0 (1, 23)
Time to resolution, days, median (min, max)	3.0 (1, 26)
Tocilizumab use, n (%)	27 (39.1)
Grade ≥3 cytopenia at Week 4 post infusion, n (%)	
Any	17 (24.6)
Anemia	7 (10.1)
Neutropenia	8 (11.6)
Thrombocytopenia	11 (15.9)
Immune effector cell-associated neurotoxicity syndrome, n (%)	
Any grade	1 (1.4)
Grade ≥3	0

Conclusions

- Pooled investigator-assessed ORR in heavily pretreated patients with synovial sarcoma or MRCLS was 36.2%, with a median duration of response of 52.0 weeks.
- Baseline tumor burden, prior systemic treatment history, and MAGE-A4 tumor expression levels are potentially important factors associated with response to afami-cel.
- A favorable benefit to risk profile was observed.
 - CRS was common but mostly low grade.
 - ICANS was uncommon.
 - Hematological toxicity was reversible with LD chemotherapy fludarabine 30 mg/m^2 x4 days and cyclophosphamide 600 mg/m^2 x3 days.
- Patients responding to afami-cel had a longer PFS than non-responders.
- These findings will inform future prognostic studies with afami-cel in soft-tissue sarcomas.

466
American Society of Clinical Oncology (ASCO), June 3–7, 2022; Hybrid, Chicago, IL, USA

References

1. Van Tine, et al. Paper 30: CTOS 2021; Virtual.

Footnotes and Abbreviations Used in Text

[a]H-score: 1 × (% of 1+ cells) + 2 × (% of 2+ cells) + 3 × (% of 3+ cells). CRS, cytokine release syndrome; HLA, human leukocyte antigen; ICANS, immune effector cell-associated neurotoxicity syndrome; LD, lymphodepletion; MAGE-A4, melanoma-associated antigen A4; mITT, modified intention-to-treat; MRCLS, myxoid/round cell liposarcoma; ORR, overall response rate; PFS, progression-free survival; RECIST, Response Evaluation Criteria in Solid Tumors.

Acknowledgements and Disclosures

Writing assistance was provided by Gabrielle Knafler, MSc, PhD, of Excel Scientific Solutions, which was contracted and compensated by Adaptimmune for these services. Sandra P. D’Angelo (dangelos@mskcc.org): Consultant for: Adaptimmune, Amgen, EMD Serono, GlaxoSmithKline, Incyte, Immune Design, Immunocore, Merck, and Nektar; Grant/Research support from: Amgen, Bristol-Myers Squibb, Deciphera, EMD Serono, Incyte, Merck, and Nektar; Travel, Accommodations, Expenses from: Adaptimmune, EMD Serono, and Nektar.