Oral Administration of Amphotericin B: Toxicokinetic Studies in Animal Models

Suraj Kalbag, Ruying Lu, Joel Ngoje, and Raphael J. Mannino

Abstract

Purpose of study: Cochleates are solid, multilayered particles of anhydrous lipid crystals capable of encapsulating hydrophobic drugs like Amphotericin B (AmB) without covalent bonding. Encochleated Amphotericin B (CAmB) is a suspension formulation of Amphotericin B cochleates currently under evaluation as an orally administered treatment against invasive fungal infections. A 28-day toxicity study was conducted to determine potential toxic effects, target organs of toxicity, and a no observable adverse effect level (NOAEL) in rats and dogs following daily oral dosing with CAmB.
Description of study: Large scale GLP batches of CAmB and placebo (cochleate vehicle without AmB) were prepared for these 28 day studies. CAmB was administered as single daily doses for 28 days by oral gavage to 10 Beagle dogs per group (5 m, 5 f) at doses of 15, 30, and 45 mg/kg and to 48 Sprague-Dawley rats per group (24 m 24 f) at doses of 30, 45, and 90 mg/kg. In rats, 18 animals (9 m, 9 f) per dose group served as a satellite group for toxicokinetic (TK) samples (blood, urine, and fecal samples for drug analysis). Animals in the placebo groups received volume-matched to the highest dose of CAmB in both species. TK blood (Days 1 and 28), urine and feces (weeks 1 and 4), and tissue samples (at necropsy) were collected for AmB quantitation by validated LC-MS methods. Histopathology analysis and serum clinical chemistry tests were done on all animals.
Results: CAmB was well tolerated by rats and dogs at all doses with no mortalities or clinical abnormalities found in either species based on comparison with historical controls. Pharmacokinetic data in dogs for Day 1 were comparable to our previous 7-day study. The increase in maximum plasma AmB concentration (C_{max}) in dog was not dose-dependent after 28 days, similar to what had been observed on Day 7 in the 7-day study. In dog, the kidney had quantifiable concentrations of AmB at all doses while liver, lung, and spleen had AmB concentrations detectable at all 3 doses. In rats, quantifiable AmB concentrations were found in liver, lung, and kidney, but not in spleen. Additional analytical and histopathological analyses are underway in dog and rat.
Conclusions: This study demonstrates that single daily oral doses of CAmB administered for 28 days are well tolerated in all dose groups in the rat and dog. The NOAEL for males and females was considered to be at least 45 mg/kg for dog and 90 mg/kg for rat. Quantifiable concentrations of AmB were measured in kidney of the dog and the rat as well as the liver and lung of the rat.

Methods

- Large scale GLP batches of CAmB and placebo (cochleate vehicle without AmB) were prepared for these 28 day studies with an AmB concentration of 5 mg/mL.
- CAmB was administered daily as single doses by oral gavage to 10 dogs per group (5 m, 5 f) at doses of 15, 30, and 45 mg/kg and to 48 rats (24 m, 24 f) at doses of 30, 45, and 90 mg/kg.
- Cardiovascular parameters in dog were evaluated pre-study and in week 4.
- In rats, 18 animals (9 m, 9 f) per dose group served as a satellite group for TK samples (blood, urine, and fecal samples for drug analysis).
- Animals were sacrificed on day 29 or on Day 42, 2 weeks after last dose administration.
- TK blood (Days 1 and 28 (dog) and 23 (rat)), urine and feces (weeks 1 and 4), and tissue samples (at necropsy) were collected for AmB quantitation by validated LC-MS methods.
- Clinical pathology, serum chemistry tests, urinalysis, and histopathology analysis were done on all animals.

Preliminary Results

Mortality/Morbidity and Clinical Observations
- All animals survived until their scheduled necropsy, except two female dogs in the high dose group. Microscopic evaluation confirmed that the deaths of the two dogs were not related to the test article but to errors in gavage administration. No clinical changes of toxicological significance were found in the control or BAmB-treated groups.

Body Weights
- No meaningful differences in body weight and body weight gain between control and BAmB-treated groups were observed.

Food Consumption
- No statistically significant changes in food consumption in control versus treated groups.

Body Temperature
- Changes in body temperature (females in treated groups, Day 28) were within normal range.

Clinical Pathology Evaluations

Serum Chemistry and Coagulation
- All changes at pre-study and post-dose administration were sporadic, slight, and within their respective normal reference ranges and were not considered to be treatment-related. Importantly, in rats and dogs, creatinine and blood urea nitrogen were within normal ranges.

Urinalysis
- No differences between control and BAmB-treated groups.

Cardiology
- There were no electrocardiographic abnormalities or blood pressure changes related to oral administration of CAmB.

Histopathology
- Any microscopic findings for dogs surviving until study termination were considered unrelated to the administration of the vehicle (controls) or CAmB. The findings were considered related to spontaneous disease or conditions, or to procedures related to necropsy activities.

Plasma and Urine Drug Levels
- In dogs, plasma levels of AmB were below the lower limit of quantitation (LLOQ) of 20 ng/mL at several time-points following the administration of low dose (15 mg/kg), especially on Day 1. Overall, plasma drug levels on Day 28 were higher than those on Day 1 at all 3 doses of CAmB. All urine concentrations were above the LLOQ of 20 ng/mL.

Urinary Excretion
- Urinary excretion over 24 hr on Days 1 and 28 showed no clear trend of drug accumulation in urine in contrast to plasma. AmB excretion in urine was negligible, ranging from 0.02 to 0.05% of dose administered.

Preliminary Toxicokinetic Analysis

- The mean t_{max} values from all dose groups ranged from 7.2 to 17.6 hr on Day 1 and from 3.6 to 8.4 hr on Day 28.
- There appeared to be an accumulation of AmB with repeat dose administration, as indicated by increased C_{max} and AUC_{last} values on Day 28 when compared with corresponding values on Day 1. The mean C_{max} values on Day 28 were ~ 1.1- to 2.3-fold higher than those obtained for Day 1. Likewise, the mean AUC_{last} estimates on Day 28 were ~ 1.9- to 7.5-fold higher on Day 28 than those estimated for Day 1.
- Plasma $t_{1/2}$ could not be estimated for several animals, especially at the lowest dose and on Day 1, due to the limited number of plasma concentrations after t_{max} that were > LLOQ of the bioanalytical assay, or due to the lack of a good fit (r < 0.8) of the best-fit line in the terminal elimination phase.
- There appeared to be no major differences in the plasma toxicokinetics of AmB between males and females on both days of blood sampling.

Toxicokinetic Parameters of Amphotericin B on Day 1 and Day 28 after Oral Administration of CAmB in Male and Female Dogs

	Day 1				Day 28			
Dose/ Gender	t_{max} (h) Mean (SD)	C_{max} (ng/mL) Mean (SD)	AUC_{last} (ng.h/mL) Mean (SD)	$t_{1/2}$ (h) Mean (SD)	t_{max} (h) Mean (SD)	C_{max} (ng/mL) Mean (SD)	AUC_{last} (ng.h/mL) Mean (SD)	$t_{1/2}$ (h) Mean (SD)
15 mg/kg								
Males	8.0 (8.9)	51.9 (20.0)	304.6 (117.6)	NC[a]	6.6 (9.8)	118.2 (20.6)	2295.4 (701.6)	32.0 (26.5)
Females	7.2 (1.8)	67.3 (36.1)	887.2 (678.1)	NC[a]	8.4 (9.0)	131.2 (35.6)	1953.6 (879.2)	10.5 (NA[b])
30 mg/kg								
Males	14.4 (8.8)	80.4 (26.9)	1389.4 (596.8)	14.0 (NA[b])	3.6 (4.0)	187.3 (52.4)	3311.5 (1074.1)	13.0 (NA[b])
Females	17.6 (8.8)	81.8 (14.6)	1299.6 (161.2)	NC[a]	8.0 (8.9)	146.1 (55.5)	2521.8 (881.3)	29.8 (29.6)
45 mg/kg								
Males	14.4 (8.8)	114.5 (51.9)	2062.4 (919.9)	59.0 (NA[b])	4.9 (3.2)	198.8 (96.1)	3865.7 (1606.7)	83.0 (NA[b])
Females	13.6 (9.6)	67.5 (10.9)	1242.4 (113.1)	28.5 (NA[b])	4.3 (2.9)	155.0 (26.1)	2836.4 (376.3)	40.5 (20.5)

[a] NC = not calculated. $t_{1/2}$ values could not be calculated for some animals due to limited number of plasma concentrations after t_{max} that were > LLOQ or r-value of best-fit line was < 0.8.
[b] NA = not applicable. $t_{1/2}$ was calculated for only 1 or 2 animals in these groups.

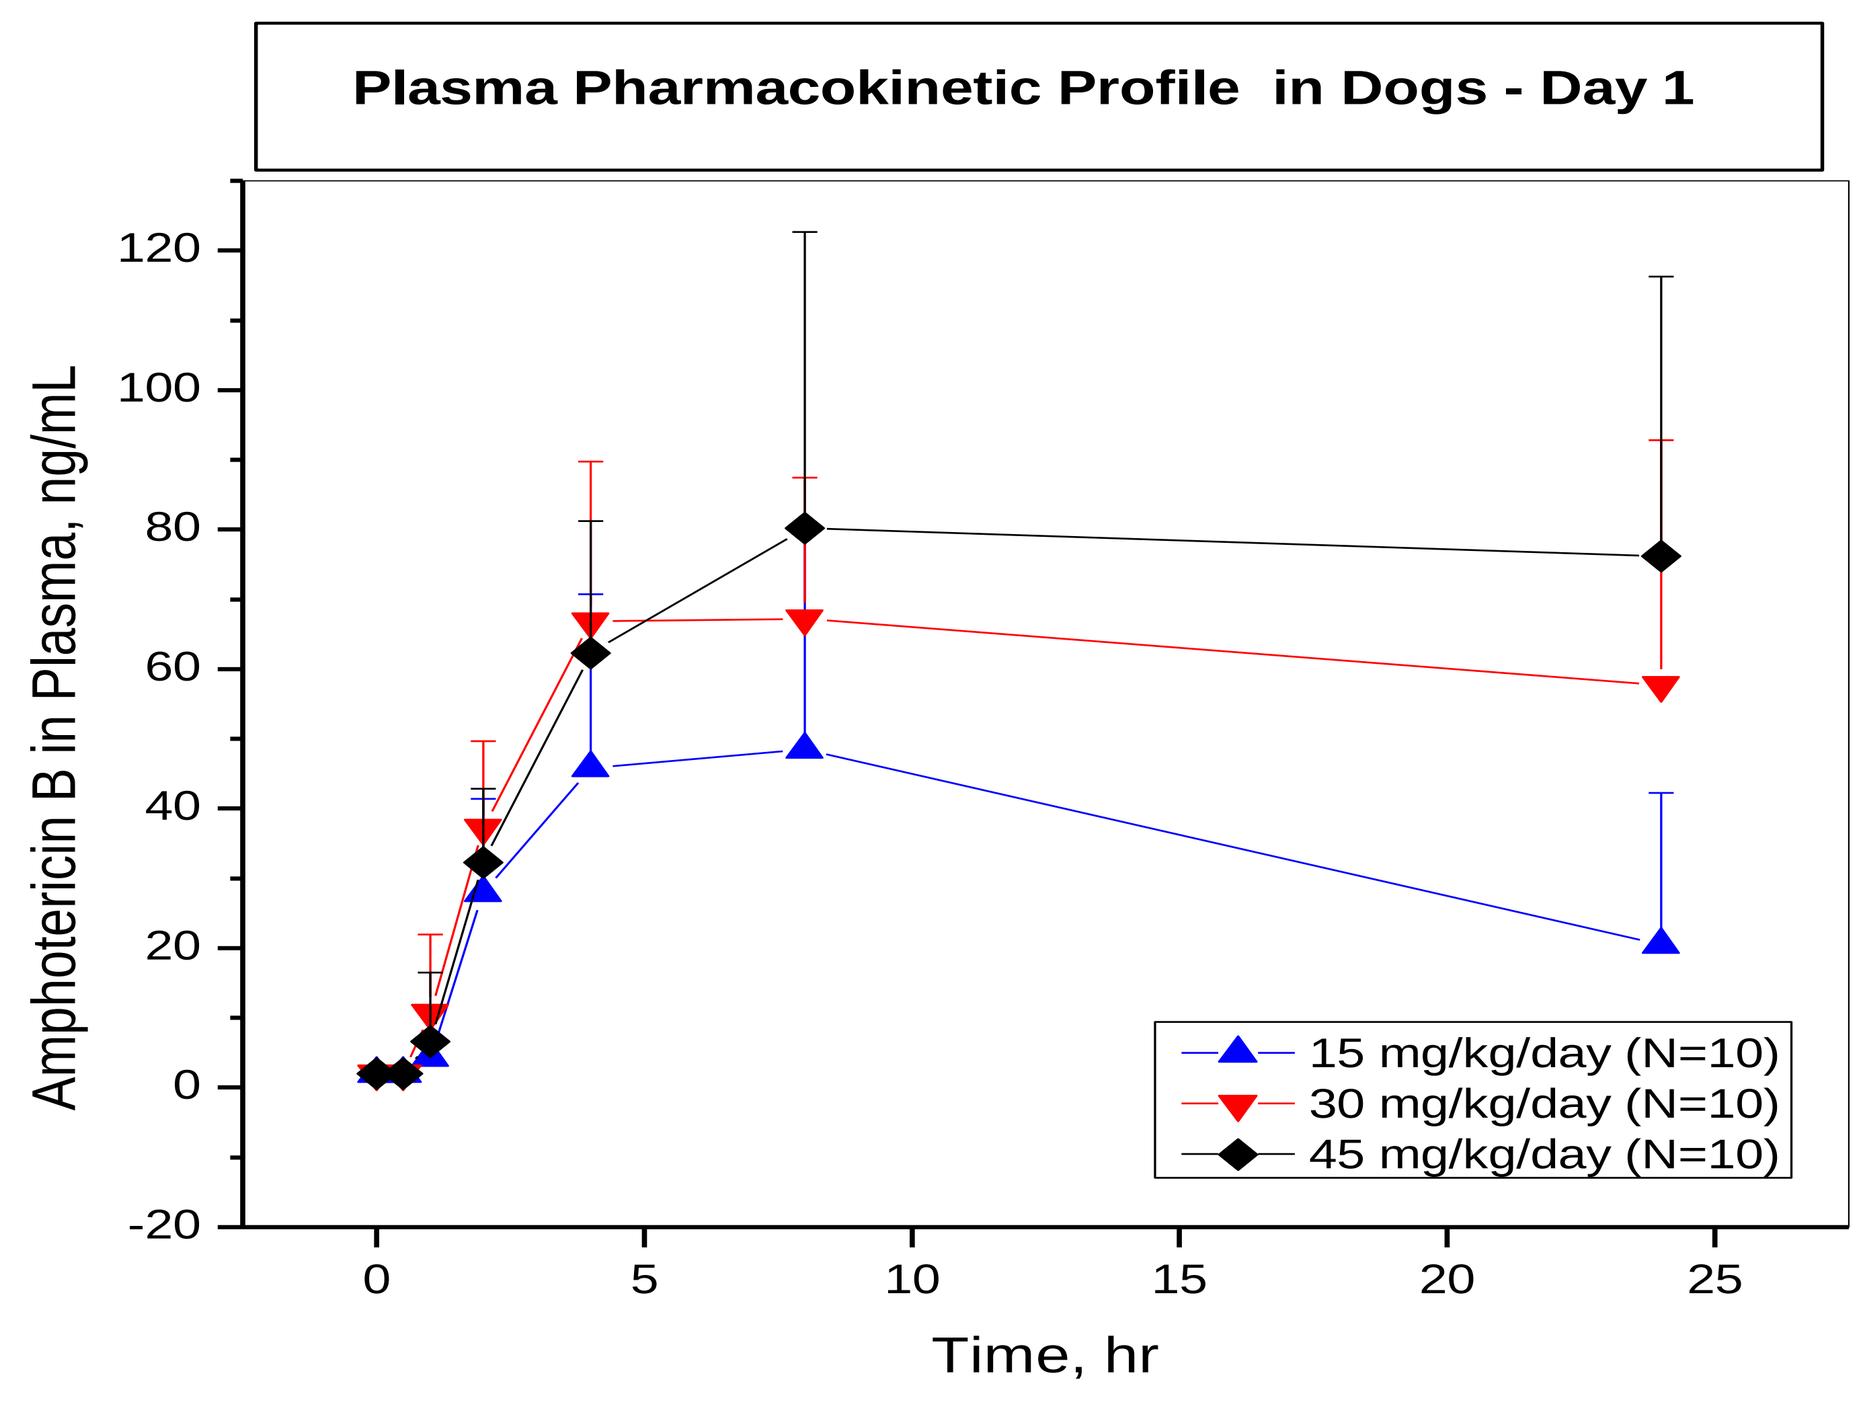
Plasma Pharmacokinetic Profile in Dogs - Day 1
Amphotericin B in Plasma, ng/mL
Time, hr
15 mg/kg/day (N=10)
30 mg/kg/day (N=10)
45 mg/kg/day (N=10)

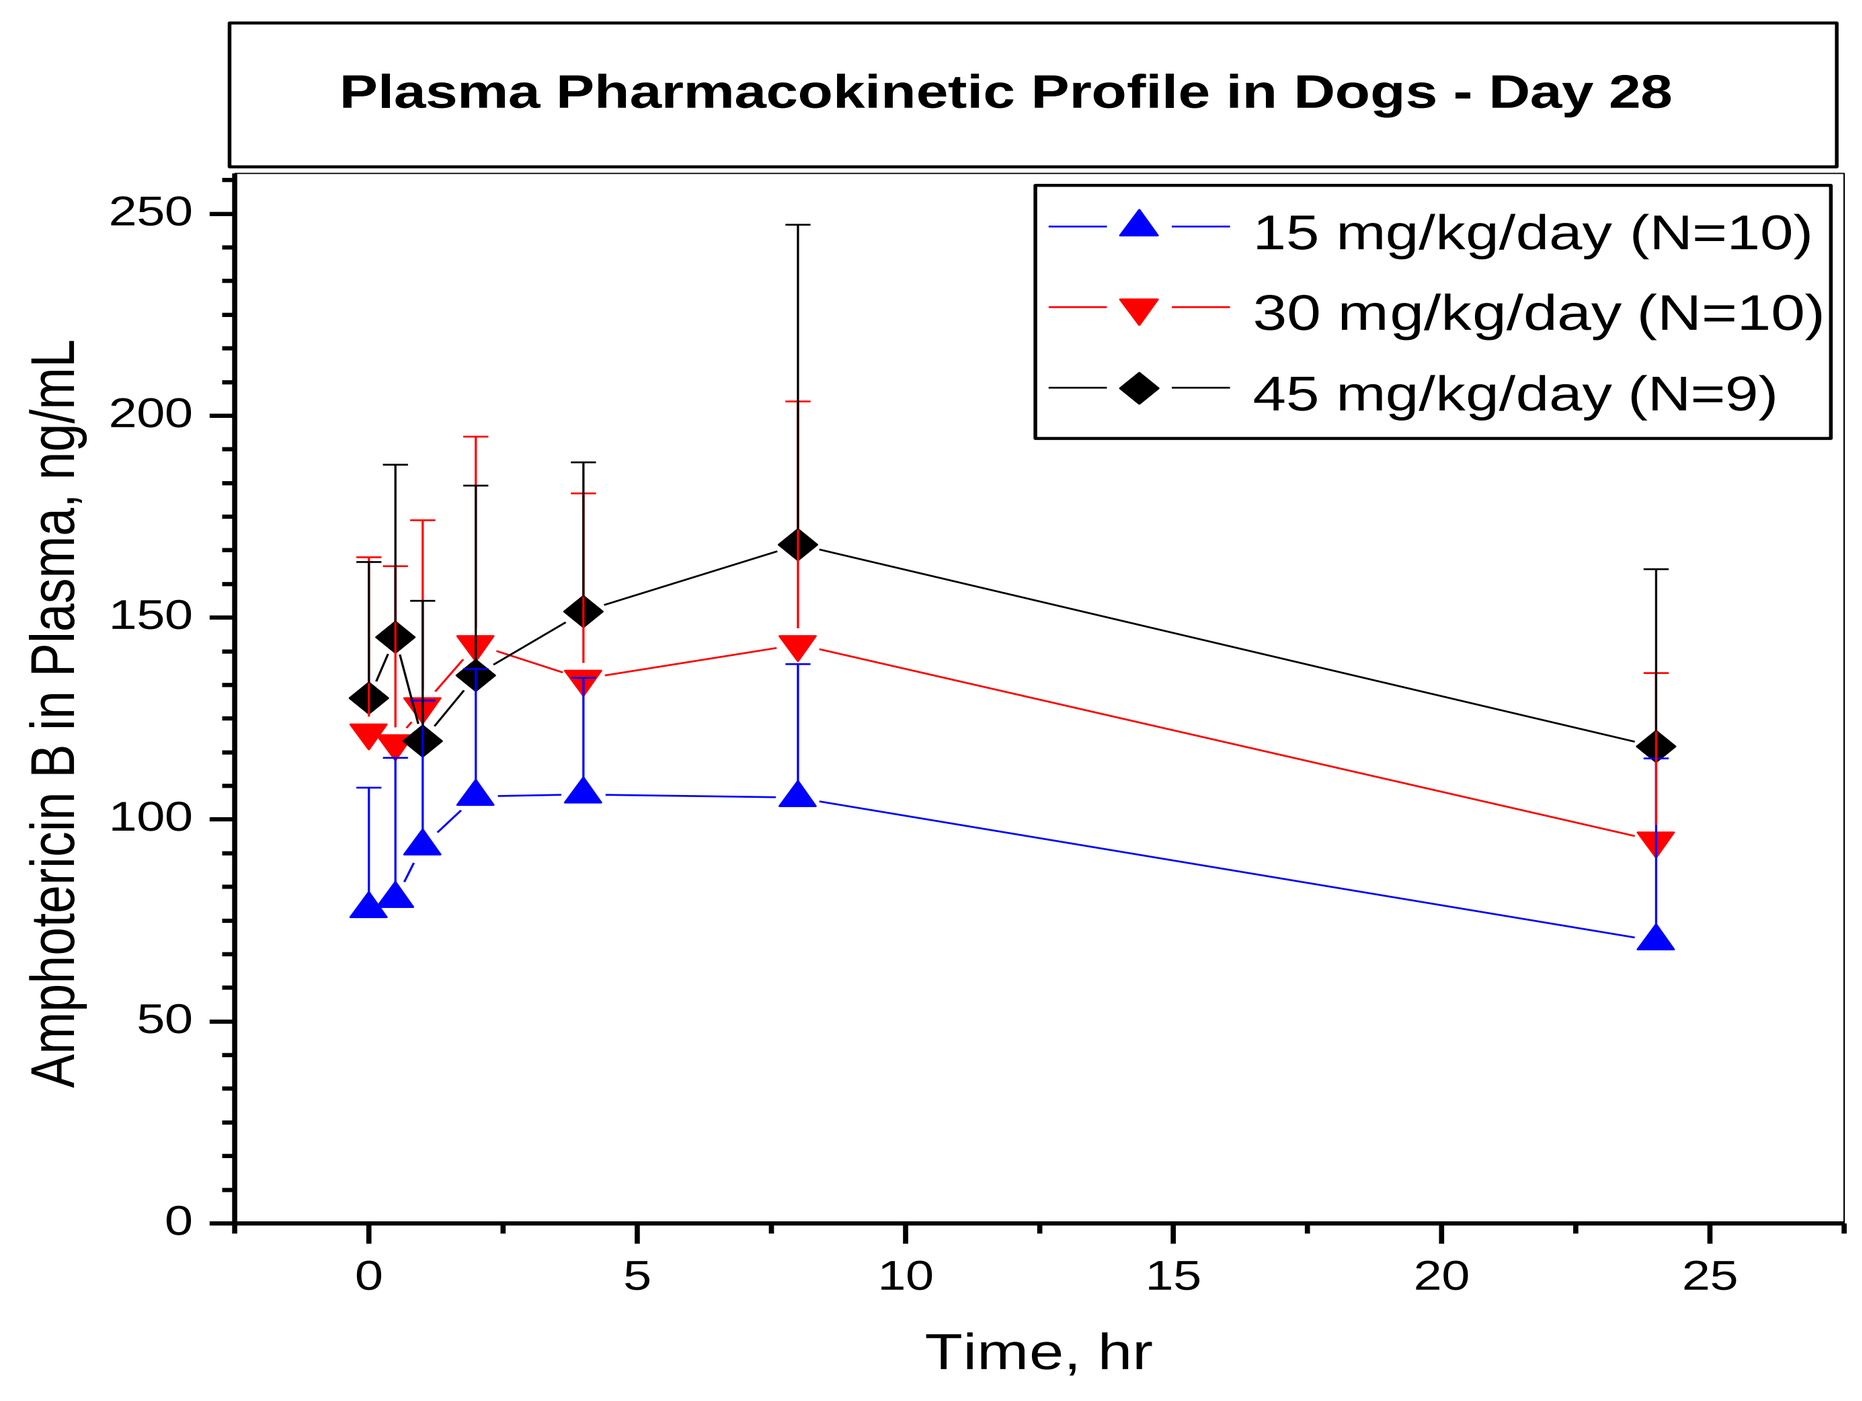
Plasma Pharmacokinetic Profile in Dogs - Day 28
Amphotericin B in Plasma, ng/mL
Time, hr
15 mg/kg/day (N=10)
30 mg/kg/day (N=10)
45 mg/kg/day (N=9)

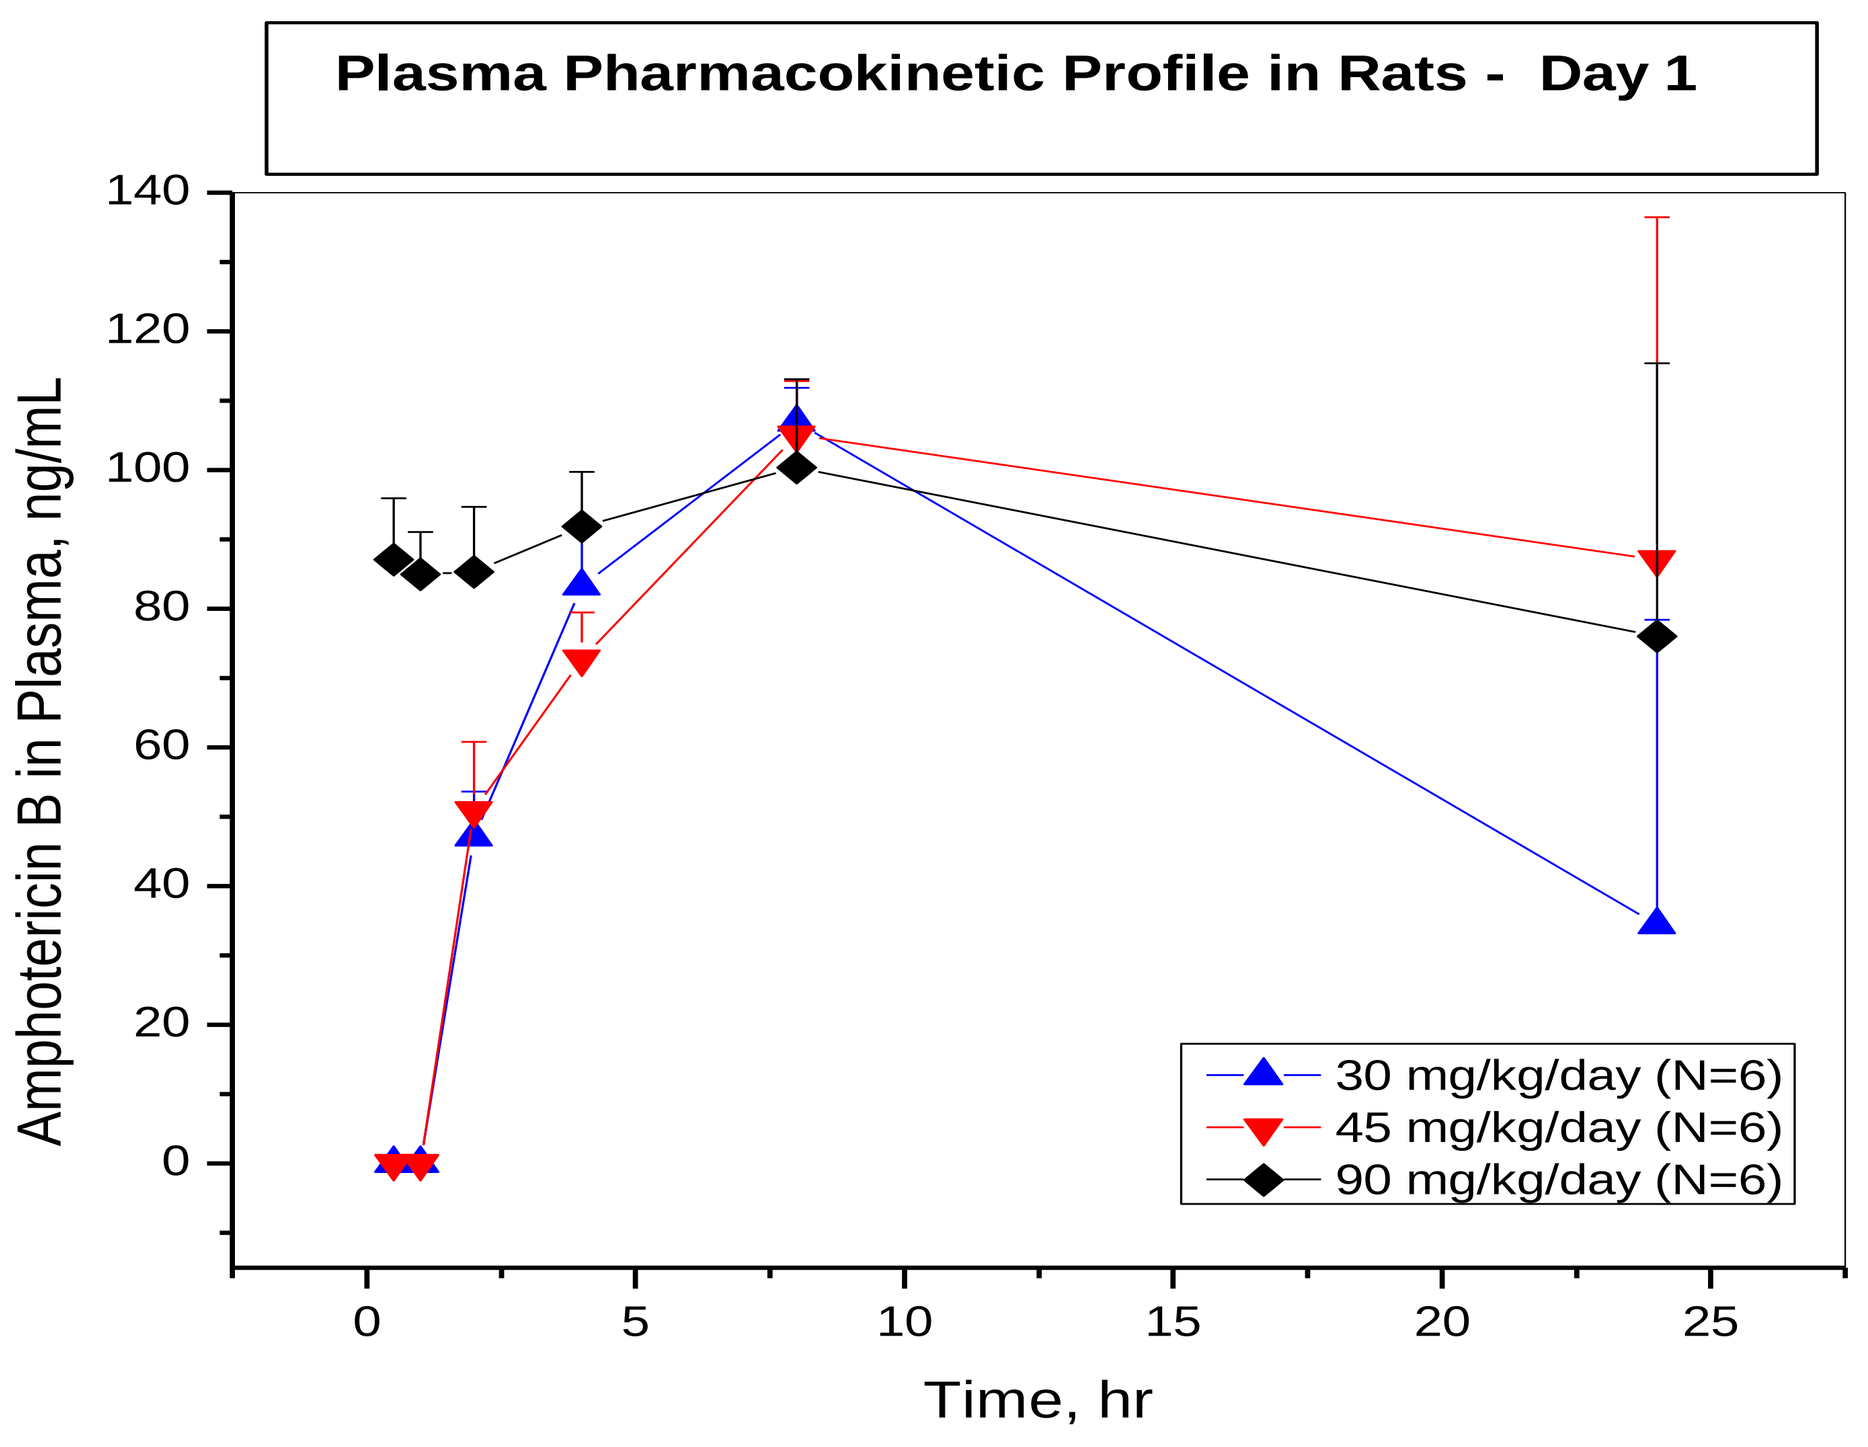
Plasma Pharmacokinetic Profile in Rats - Day 1
Amphotericin B in Plasma, ng/mL
Time, hr
30 mg/kg/day (N=6)
45 mg/kg/day (N=6)
90 mg/kg/day (N=6)

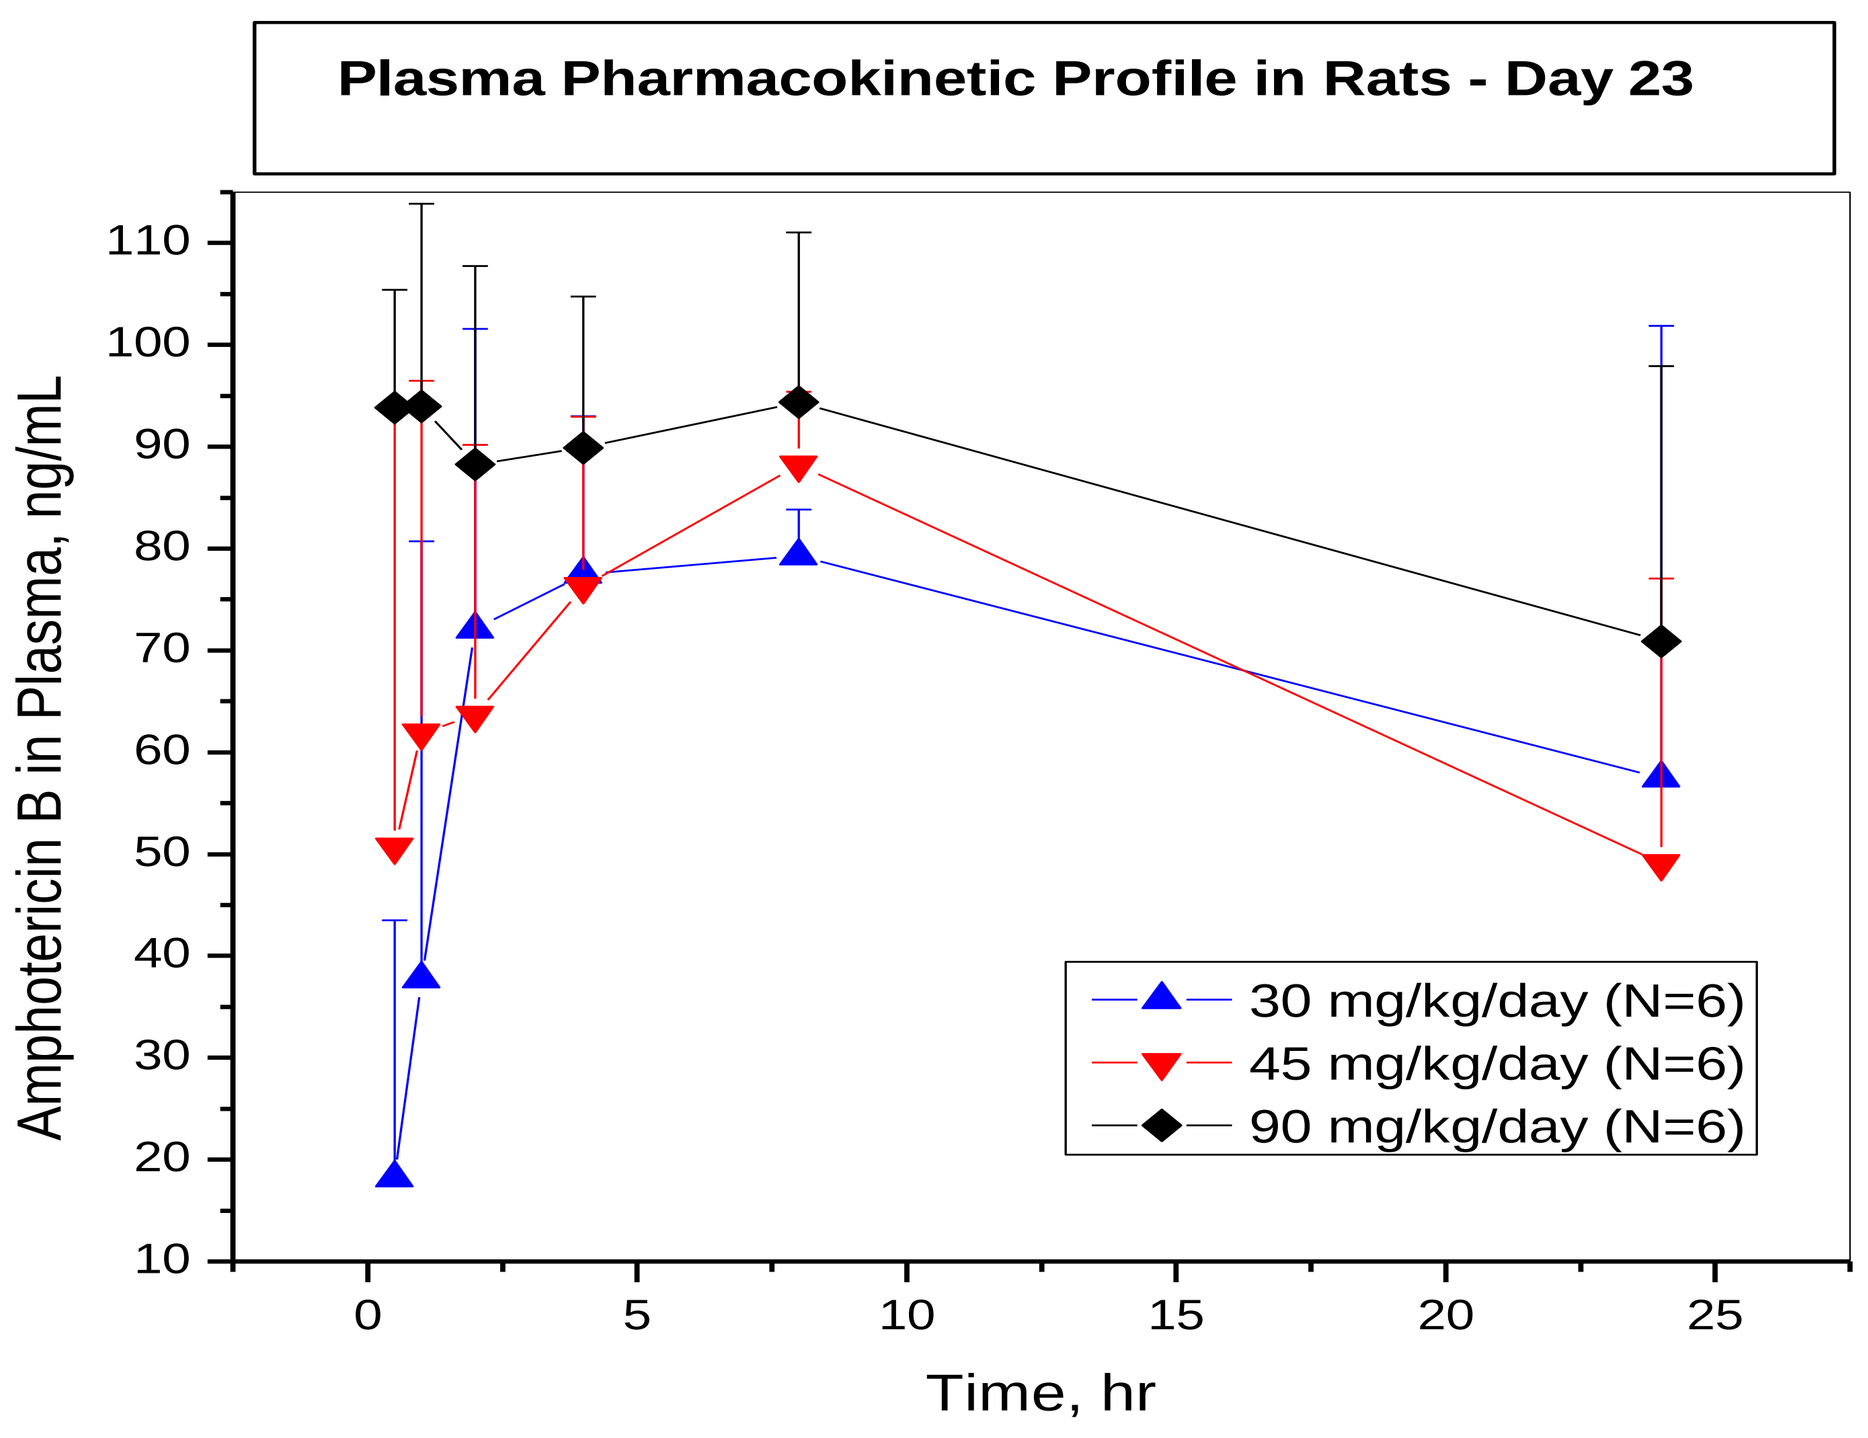
Plasma Pharmacokinetic Profile in Rats - Day 23
Amphotericin B in Plasma, ng/mL
Time, hr
30 mg/kg/day (N=6)
45 mg/kg/day (N=6)
90 mg/kg/day (N=6)

Preliminary Conclusions

- Daily oral administration of CAmB to male and female beagle dogs at 15, 30, or 45 mg/kg/day and male and female rats at 30, 45, or 90 mg/kg/day did not produce overt adverse effects.
- The NOAEL is at least 45 mg/kg/day in dogs and 90 mg/kg/day in rats for daily oral dose administration of CAmB for 28 consecutive days.
- No test article-related effects were seen in the following parameters: clinical observations, body weights, food consumption, body temperature, clinical pathology (hematology, serum chemistry and coagulation), urinalysis, ophthalmology, cardiology, organ weights, macroscopic and microscopic evaluations.
- Toxicokinetic analysis indicated an accumulation of AmB in all 3 dose groups on Day 28 after repeat administration as indicated by increased C_{max} and AUC_{last}.
- Urinary excretion of drug was negligible when compared with the actual dose administered. There were no major differences between male and female dogs in the disposition of drug at the dose levels studied.
- Tissue AmB analysis showed quantifiable concentrations in dog kidney but only detectable levels in liver, lung, and spleen. In the rat, the liver, lung, and kidney showed quantifiable AmB concentrations but not in spleen.

We thank Drs. Chris Lambros and Hao Zhang (Division of AIDS, NIAID) for their continued support.