SEVERE ORAL MUCOSITIS (SOM) MITIGATION BY GENETICALLY MODIFIED LACTOCOCCUS LACTIS BACTERIA (LLB) PRODUCING HUMAN TREFOIL FACTOR 1 (hTFF1; AG013) IN PATIENTS BEING TREATED WITH CONCOMITANT CHEMORADIATION (CRT) FOR ORAL AND OROPHARYNGEAL CANCERS (OCOPC)

Suraj Singh[1], Sandra Nuyts[2], Robert Doline[3], Suma Satti[4], Matthew Schwartz[5], Stephen Thatcher[6], Yuhchyau Chen[7], Sanford Katz[8], James Wade[9], Madhur Garg[10], Jill Wagemans[11], Pol Specenier[12], Claus Wittekindt[13], Lip Lee[14], John Riefler[15], Stephen Sonis[16], Michael Emanuel[17], Fiona Cilli[17], Alan Joslyn[17]

ABSTRACT

BACKGROUND: SOM is a devasting consequence of CRT. hTFF1 is a naturally occurring protein that can protect the mucosa. LLB were genetically modified (GM) to produce hTFF1, formulated as an oral rinse and attenuated SOM in patients receiving chemotherapy. The GM LLB lack all necessary components for survival and multiplication. The objective of this ongoing Phase 2 trial is to assess the safety and efficacy of AG013 as a SOM intervention.

METHODS: This is a double-blind, randomized, placebo-controlled trial recruiting ~200 patients with OCOPC at 48 sites (US and Europe). Patients (PTS) receive cumulative RT (cumRT) between 50 Gy – 72 Gy, 2.0-2.2 Gy QD + QW/Q3W cisplatin. At least two mucosal sites at risk of SOM receive minimal cumRT of 50 Gy. PTS are randomized 1:1 to receive placebo or AG013 [LLB strain sAGX0085 engineered to secrete hTFF1 (2x1011 CFU/15 mL tid)] starting on CRT day 1 and continuing 2 weeks post-CRT. Beginning on CRT day 1 and continuing Q2W until resolution, OM is assessed by trained assessors and scores assigned centrally. The primary and secondary efficacy endpoints are SOM duration and incidence (WHO criteria). AEs are described by NCI-CTCv4. A DSMB performed a safety analysis following accrual of the first 24 PTS. Tumor response to CRT is evaluated for 1-year post CRT.

RESULTS: 71 PTS have been randomized across 48 study sites. Complete OM data are available for 42 PTS for whom blinded evaluation (active and placebo) demonstrated an overall SOM incidence of 52%. SOM was noted at 81 of 547 visits (14.8%). 25 PTS have stopped active treatment; 2 for non-compliance, 5 for AEs, and 18 lost to follow-up or unwilling or unable to conform to the protocol. Unexpected SAEs were noted in 9 PTS. No study drug-associated cases of bacteremia or sepsis were seen. DSMB review after the first 24 PTS concluded that there were no contraindications to study continuance.

CONCLUSIONS: Observations based on blinded data suggest that AG013 offers a safe, well-tolerated, and potentially efficacious platform to deliver an effective protein intervention for SOM mitigation.

METHODS

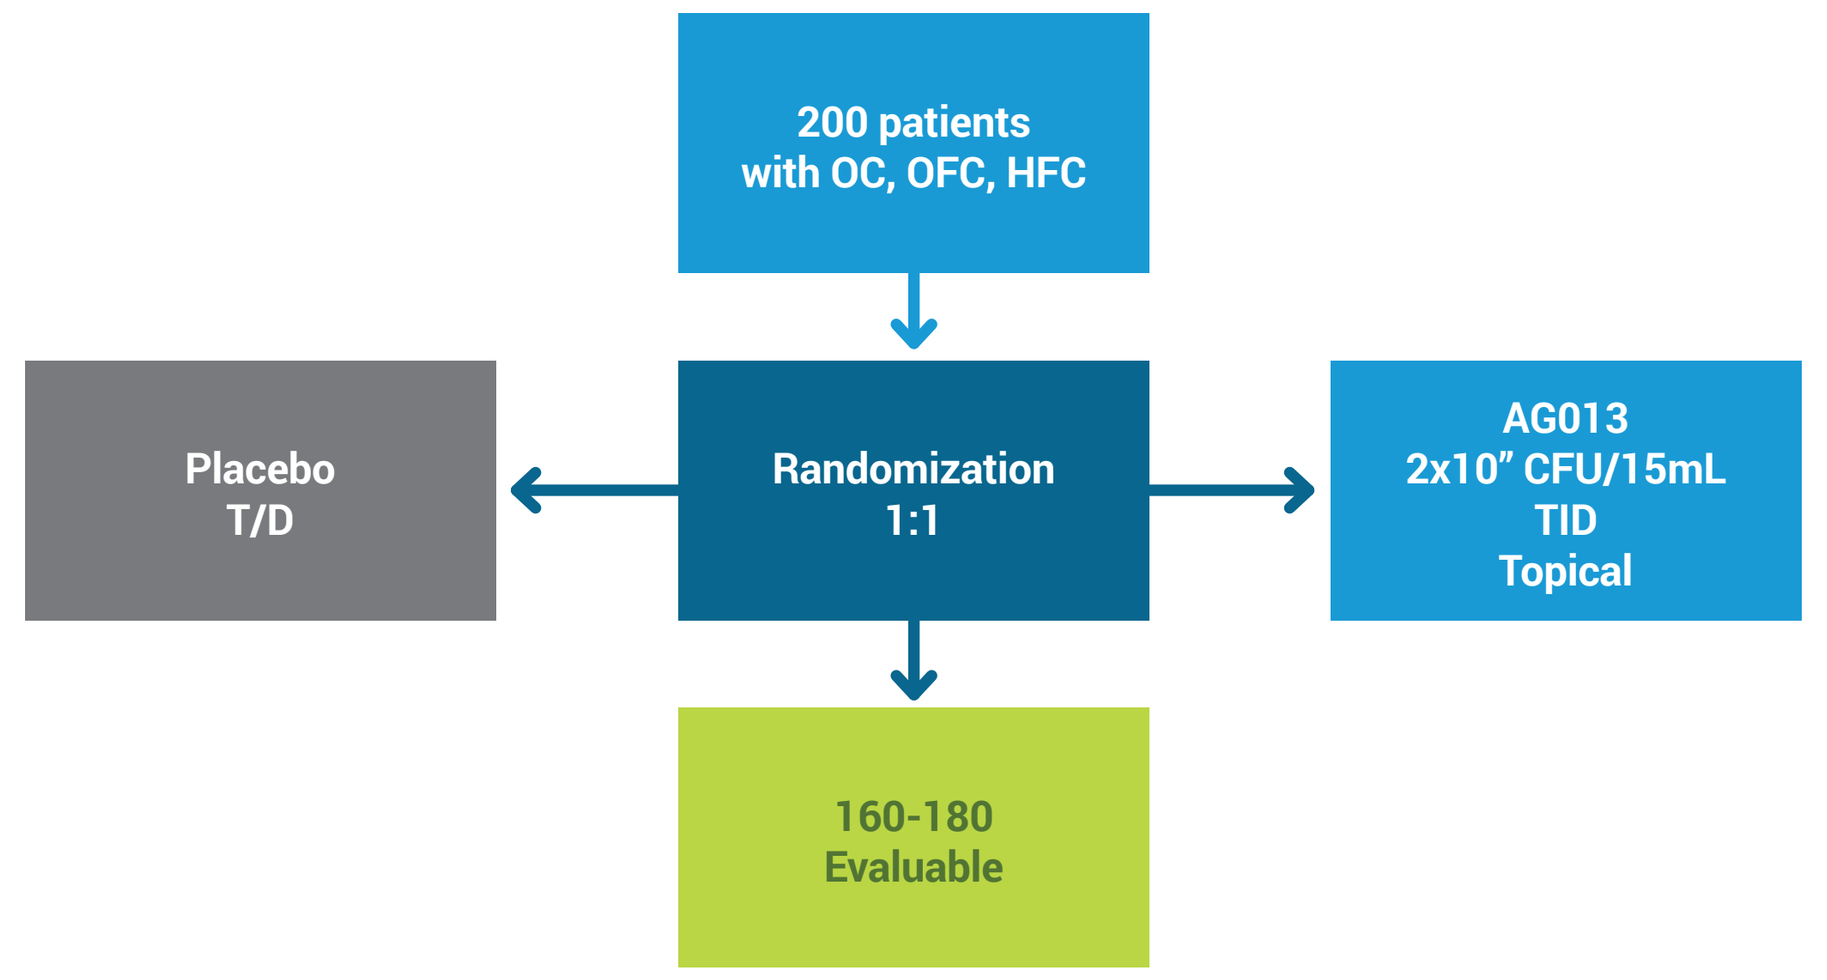
200 patients
with OC, OFC, HFC
Placebo
T/D
Randomization
1:1
AG013
2x10" CFU/15mL
TID
Topical
160-180
Evaluable

Primary efficacy endpoint:
Duration (in days) of severe OM (WHO grades 3 (unable to eat) & 4 (unable to drink))

Sample size consideration:
160 evaluable patients (80/group) provides 80% power to detect 5-day difference between groups with respect to severe OM

OM secondary endpoints:
Number of OM free (WHO grades 1 & 2) days, time to onset, use of pain medication, alteration in cancer regimens; emergency room visits for SOM

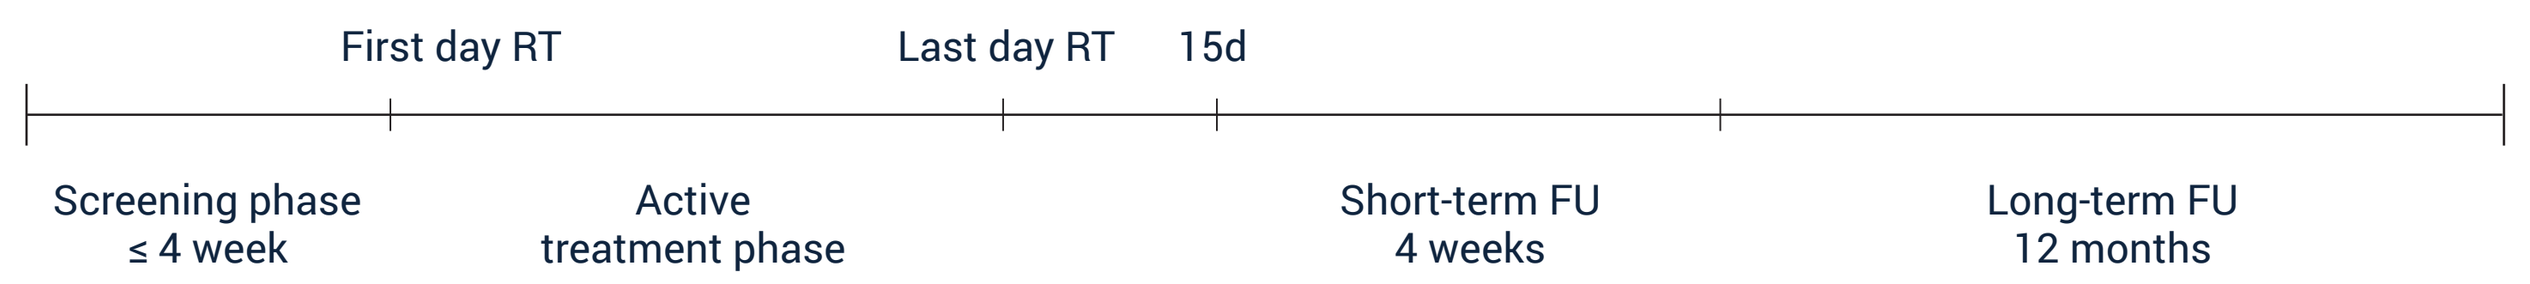
First day RT
Last day RT
15d
Screening phase
≤ 4 week
Active
treatment phase
Short-term FU
4 weeks
Long-term FU
12 months

UPDATED GLOBAL ENROLMENT STATUS

Patient Status	Total/Actual
Pre-screened pending	5 USA + 1 GER + 1 BEL + 2 UK
Screened	189
Currently in screening	9
Screen failure	30 (15.87%)
Enrolled	151 (75.5)%
Randomized & treated	72
Completed treatment	50

	Country Enrolment
Belgium	10
Germany	25
UK	5
USA	111
TOTAL	151

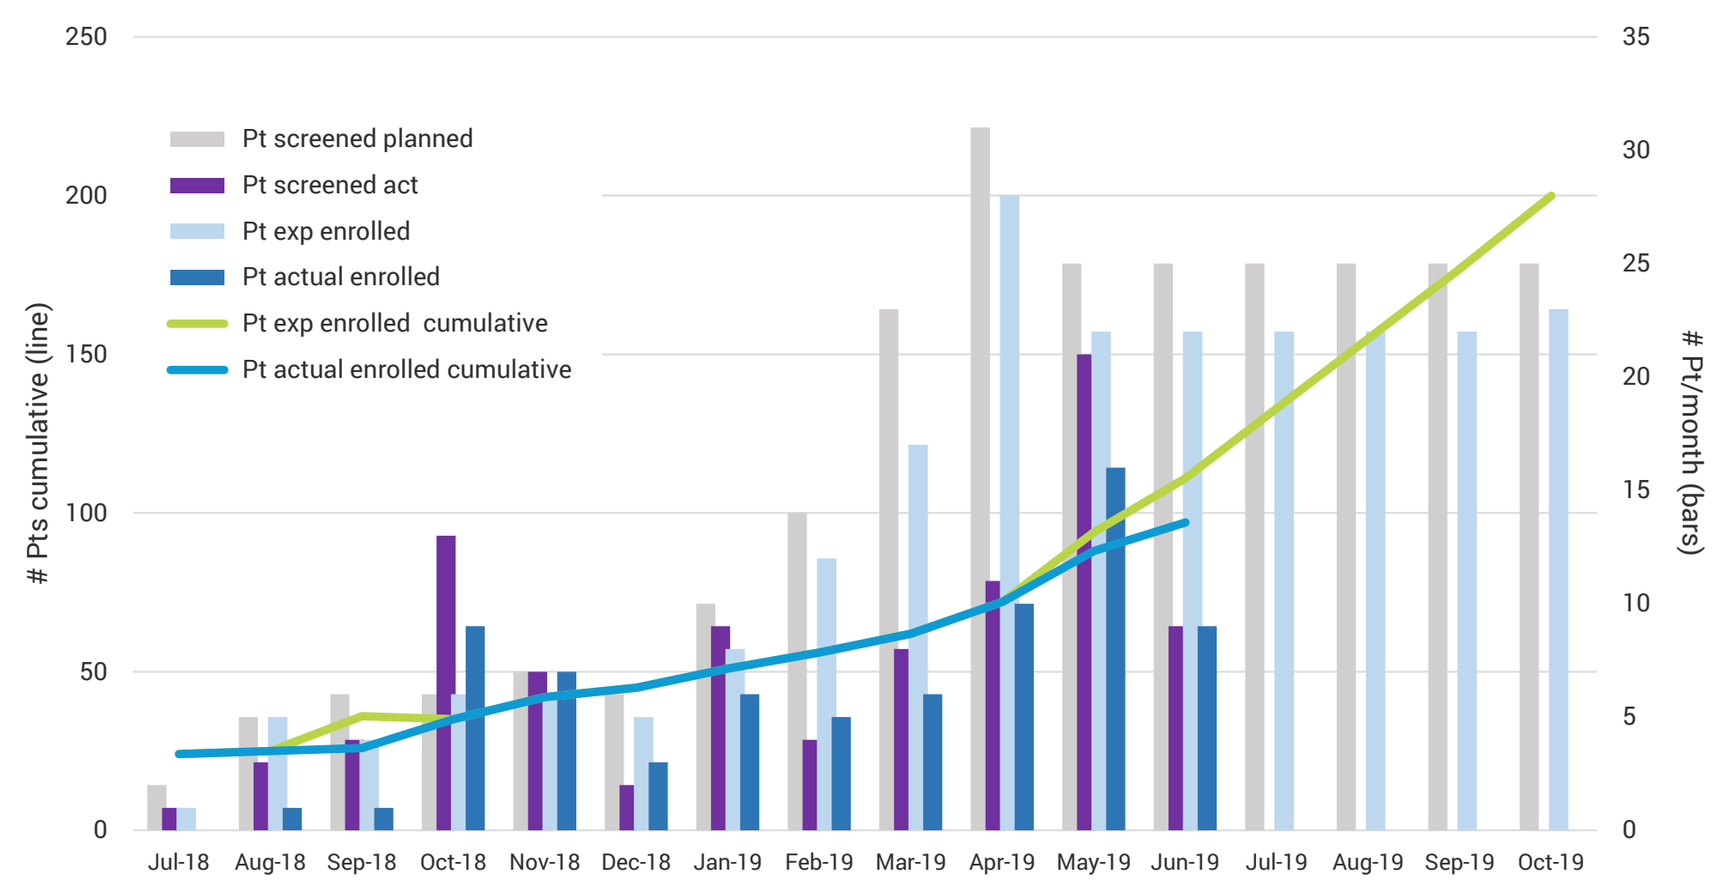
Pt screened planned
Pt screened act
Pt exp enrolled
Pt actual enrolled
Pt exp enrolled cumulative
Pt actual enrolled cumulative
Pts cumulative (line)
Pt/month (bars)
250
200
150
100
50
0
35
30
25
20
15
10
5
0
Jul-18
Aug-18
Sep-18
Oct-18
Nov-18
Dec-18
Jan-19
Feb-19
Mar-19
Apr-19
May-19
Jun-19
Jul-19
Aug-19
Sep-19
Oct-19

GENETICALLY MODIFIED BACTERIAL SYSTEM

- Convenient, flavored oral rinsing solution composed of genetically modified Lactococcus lactis (non-pathologic food grade bacterium) engineered to deliver mucosal protectant human Trefoil Factor 1 (hTFF1) to mucosal tissues
- Trefoil Factors (TFF's) are a class of peptides involved in protecting mucosal tissues against damage and in subsequent repair
- Cost effective (low COGs) rinse provides daily continuous oropharyngeal coverage with L. lactis producing hTFF1 during entire cancer treatment regimen

1 2 3 4 5

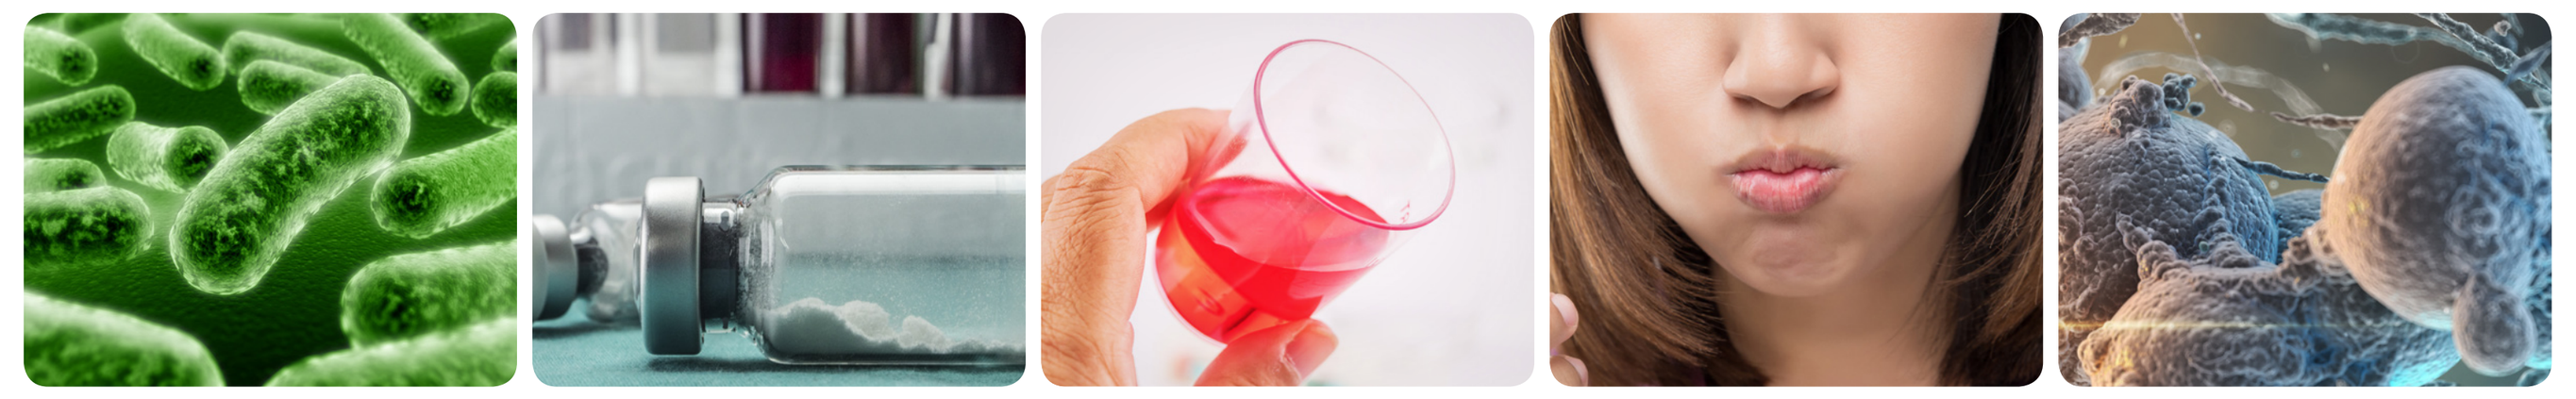

1. AG013 delivers hTTF1 via genetically modified lactococcus
2. The bacteria is freeze-dried into vials
3. Patient mixes powder with a raspberry-flavored solution
4. Patient swishes for 30 seconds after every meal
5. This activity promotes a protein called trefoil factor, which regrows the oral lining

CLINICAL STUDY SITES

United States

- **Decatur Memorial Hospital** – Decatur, IL
- **Montefiore Medical Center** – Bronx, NY
- **East Carolina University School of Dental Medicine** – Greenville, NC
- **Cancer Care Northwest** – Spokane Valley, WA
- **CaroMont Regional Medical Center** – Gastonia, NC
- **Ochsner Medical Center** – New Orleans, LA
- **MultiCare Health System** – Tacoma, WA
- **University of Michigan Medical Center** – Ann Arbor, MI
- **Columbus Regional Research Institute** – Columbus, GA
- **Willis-Knighton Cancer Center** – Shreverport, LA
- **Comprehensive Cancer Centers of Nevada-Henderson** – Henderson, NV
- **Ohio State University Comprehensive Cancer Center - Arthur G. James Cancer Hospital** – Columbus, OH
- **UPMC Shadyside Hospital** – Pittsburgh, PA
- **Orlando Health, Inc.** – Orlando, FL
- **University of Connecticut Health Center** – Farmington, CT
- **University of Rochester Medical Center** – Rochester, NY
- **Renown Regional Medical Center** – Reno, NV
- **University of Texas MD Anderson Cancer Center** – Houston, TX
- **University of Virginia Cancer Center** – Charlottesville, VA
- **Northwell Health Cancer Institute / Center for Novel Cancer Therapeutics** – Lake Success, NY
- **Norton Cancer Institute, Multidisciplinary Clinic** – Louisville, KY
- **Cardinal Bernardin Cancer Center** – Maywood, IL
- **Henry Ford Health System** – Detroit, MI
- **Helen F. Graham Cancer Center** – Newark, DE
- **Huntsman Cancer Hospital** – Salt Lake City, UT
- **Baylor Scott & White Medical Center - Temple, Vasicek Cancer Treatment Center** – Temple, TX
- **Temple University Hospital, Radiation Oncology** – Philadelphia, PA
- **St. Vincent Anderson Regional, Cancer Center** – Anderson, IN
- **Norton Cancer Institute, Multidisciplinary Clinic** – Louisville, KY

Belgium

- **University Hospitals Leuven, Campus Gasthuisberg** – Leuven
- **Jules Bordet Institute** – Brussels
- **University Hospital Antwerp (UZA)** – Edegem
- **St. Maarten General Hospital** – Mechelen
- **Charleroi Grand Hospital (GHDC)** – Charleroi
- **UCL Namur University Hospital, Site Sainte-Elisabeth** – Namur

UK

- **The Christie NHS Foundation Trust** – Manchester
- **Royal Cornwall Hospital** – Truro
- **Glasgow Beatson West of Scotland Cancer Center** – Glasgow
- **London Guy's Hospital** – London
- **Plymouth Derriford Hospital** – Devon

Germany

- **University Hospital Freiburg** – Freiburg
- **Hospital Kassel** – Kassel
- **Helios Amper Hospital** – Dachau
- **Caritas Klinikum** – Saarbruecken
- **University Hospital Regensburg** – Regensburg
- **University Hospital Schleswig-Holstein** – Kiel
- **HELIOS Hospital Krefeld** – Krefeld
- **University Hospital Giessen and Marburg GmbH** – Giessen
- **University Hospital Johannes Gutenberg - University of Mainz** – Mainz
- **University Hospital Jena, Department of Otorhinolaryngology** – Jena
- **University Hospital Aachen, Department of Otorhinology and Plastic Surgery** – Aachen
- **University Medical Center, Center for Radiology, Clinic for Radiotherapy** – Rostock
- **Hannover Medical School, Department of Radiation Oncology** – Hannover
- **University Hospital Magdeburg A.o.R., Clinic for Radiotherapy** – Magdeburg
- **University Hospital Mannheim, ENT Clinic** – GMannheim
- **Ludwig Maximilians University Hospital, Campus Grosshadern, Department of Radiotherapy and Radiation Oncology** – Munich
- **Clinics Maria Hilf GmbH - Hospital St. Franziskus, Clinic for Radiotherapy** – Moenchengladbach

BACKGROUND

- Severe oral mucositis (SOM) is a common, debilitating, treatment-disruptive, and costly side effect of concomitant chemoradiation regimens (CRT) used for the treatment of head and neck cancers.
- There is no approved intervention for the prevention and/or treatment of CRT-related SOM.
- Human trefoil factor (hTFF) is a naturally occurring protein which protects the gastrointestinal mucosa from chemotherapy- and radiotherapy-induced injury (Beck PL et al. Gastroenterology 2004; 126:796-808).
- AG013 provides a topically applied formulation in which genetically modified non-pathogenic bacteria (Lactococcus lactis) is programmed to produce therapeutic levels of hTFF1 to mucosal tissues.
- Results of a Phase 1b clinical trial of AG013 in patients receiving stomatotoxic chemotherapy-based induction therapy demonstrated a reduction in mucositis duration and use of mucositis-related health-resources (Limaye SA et al. Cancer 2013; 119:4268-76).
- Objective: The objective of this ongoing Phase 2 trial is to assess the safety and efficacy of AG013 in mitigating severe oral mucositis in patients receiving concomitant chemoradiation for the treatment of oral and oropharyngeal cancers.

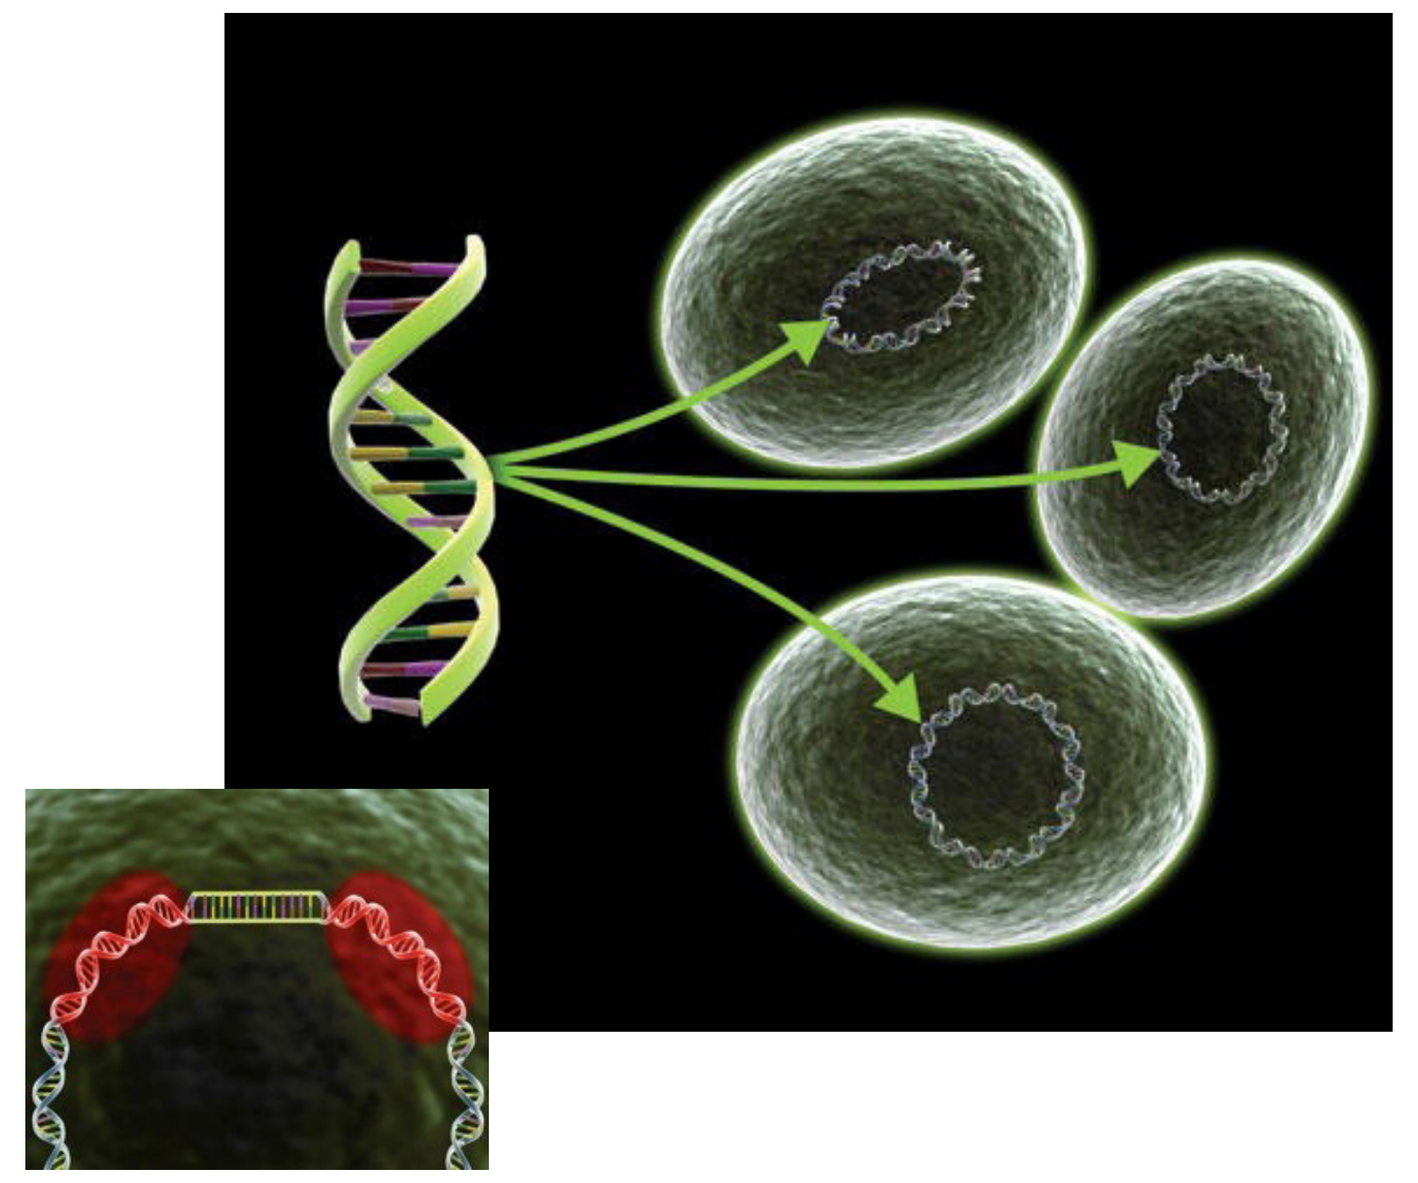

EFFICACY AND DISCONTINUATIONS

- Evaluation of blinded data which included any patient with SOM after week 1 and those who received a cumulative dose of 55 Gy (week 6) demonstrated an overall SOM incidence of 47%
- SOM was noted in 110 of 842 visits (13.1%) evaluable visits
- 41 of 151 (27%) have discontinued:
 - 6: protocol non-compliance
 - 7: adverse events
 - 5: administrative reasons
 - 23: withdrew consent including lack of efficacy

Adverse Event	% of Adverse Events
Anaemia	5%
WBC count decreased	6%
Sepsis	0% *
Anorexia	5%
Fatigue	14%
Confusion	1%
Abdominal distention	0%
Abdominal pain	0%
Constipation	12%
Nausea	22%**
Diarrhea	7%
Dysguesia	10%
Dyspepsia	8%
Odynophagia	2%
Acute kidney disease	2%
Cough	4%
Acute pulmonary embolism	0%

* Unrelated to IMP
** 17% possibly related to IMP

CONCLUSIONS

Observations based on blinded data suggest that AG013 offers a safe, well-tolerated, and potentially efficacious platform to deliver an effective protein intervention for SOM mitigation.

REFERENCES:

1. Beck PL, et al. Chemotherapy- and radiotherapy-induced intestinal damage is regulated by intestinal trefoil factor. Gastroenterology 2004; 126: 796-808.
2. Limaye SA, et al. Phase 1b, multicenter, single blinded, placebo-controlled, sequential dose escalation study to assess the safety and tolerability of topically applied AG013 in subjects with locally advanced head and neck cancer receiving induction chemotherapy. Cancer 2013; 119: 4268-76.