Proper priming of T cells is crucial for the anti-tumor effect of mPTX-35, an anti-TNFRSF25 agonist antibody

Dareen Sarhan[1], Lara Rabah[1], Vikas Tahiliani[2], Anh M. Trinh[2], Rahul R. Jasuja[2], Eric P. Dixon[2], Matthew Seavey[2], Vivek Verma[1], Seema Gupta[1], Samir N. Khleif[1]

[1]Jeannie and Tony Loop Immuno-Oncology Laboratory, Lombardi Comprehensive Cancer Center, Georgetown University School of Medicine, Washington DC, USA; [2]Pelican Therapeutics, San Antonio, TX, USA

#22-A-5397

Abstract

TNFRSF25, also known as Death Receptor 3 (DR3), is a cell surface receptor of the tumor necrosis factor receptor superfamily (TNFRSF) which is expressed on T cells, innate lymphoid cells, and B cells. Upon binding with TL1A ligand, this co-stimulatory molecule induces T cell activation, leading either to inflammation or cell death. Mouse PTX-35 (mPTX-35) is a TNFRSF25 functional agonist derived from a CDR that was humanized and affinity matured, then fused to a mouse IgG1-Fc backbone. Since costimulatory agonist antibodies have shown limited efficacy in clinical trials, our aims were to (i) determine the anti-tumor efficacy of mPTX-35, (ii) elucidate any mechanisms of resistance, and (iii) identify immune conditions that provide effective anti-tumor immune responses to mPTX-35 treatment. We studied the antitumor immune effects of mPTX-35 in both cold and hot tumor models. In a TC-1 cold tumor, we found that treatment with an anti-TNFRSF25 agonist antibody does not affect tumor growth or survival. In contrast, vaccination with an E7 antigen-specific vaccine leads to an environment that reverses primary resistance to the agonist antibody, inhibiting the tumor growth, and enhancing overall survival. Interestingly, we found that similar to other co-stimulatory agonist molecules, the immune effects of the anti-TNFRSF25 agonist are dose-dependent with lower (0.2 mg/Kg) and higher (5 mg/Kg) doses of mPTX-35 showing a reduced anti-tumor response than a dose of 1 mg/Kg, resulting in a bell-shaped response in all tumor models. Similar immune-mediated anti-tumor effects of mPTX-35 were found in a CT26 (hot) tumor model. In conclusion, these results show that anti-TNFRSF25 agonist antibody demonstrates high anti-tumor efficacy in tumors that are infiltrated with T cells while the absence of properly antigen activated CD8 T cells (such as in cold tumors) incurs primary resistance to TNFRSF25 agonist that is reversed by proper priming of CD8 T cells. These results highlight the importance of optimal priming to limit primary resistance mechanisms and drive an efficacious immune response *via* the TNSFR25 axis.

Aims

(i) Determine the anti-tumor efficacy of mPTX-35;
(ii) Elucidate any mechanisms of resistance; and
(iii) Identify immune conditions that provide effective anti-tumor immune responses to mPTX-35 treatment.

Experimental methods

Mice: C57BL/6J and BALB/c mice were used as per experimental requirements.

Tumor cell lines: TC-1 (mouse lung epithelial cells transfected with human papilloma virus encoded E6/E7 early proteins and harboring H-ras mutation) were provided by Dr. TC Wu. CT26 (murine colorectal carcinoma) cells were obtained from ATCC. Mice were inoculated subcutaneously with 7 x 10^4 TC-1 or 0.1 x 10^6 CT26 cells for tumor induction.

Vaccine: TC-1 tumor-bearing mice were vaccinated with E7 peptide. Vaccination was started when tumors reached average size of 0.08 - 0.1 cm^3. Three vaccinations at one week interval were given by subcutaneous inoculation of 100 µg of respective peptide (E7) given along with 20 µg of pan-HLA DR-binding epitope (PADRE) and 10 µg of QuilA as an adjuvant.

mPTX-35: Anti-TNFRSF25 agonist antibody (Pelican Therapeutics, Inc, San Antonio, TX) was used in the present study. mPTX-35 was given in three different schedules, before the vaccine when tumors reached average size of 0.05 - 0.06 cm^3, with the vaccine and 2 days after the vaccine.

Results

TNFRSF25 agonist induces dose-dependent bell-shaped anti-tumor response in the cold TC-1 tumor model

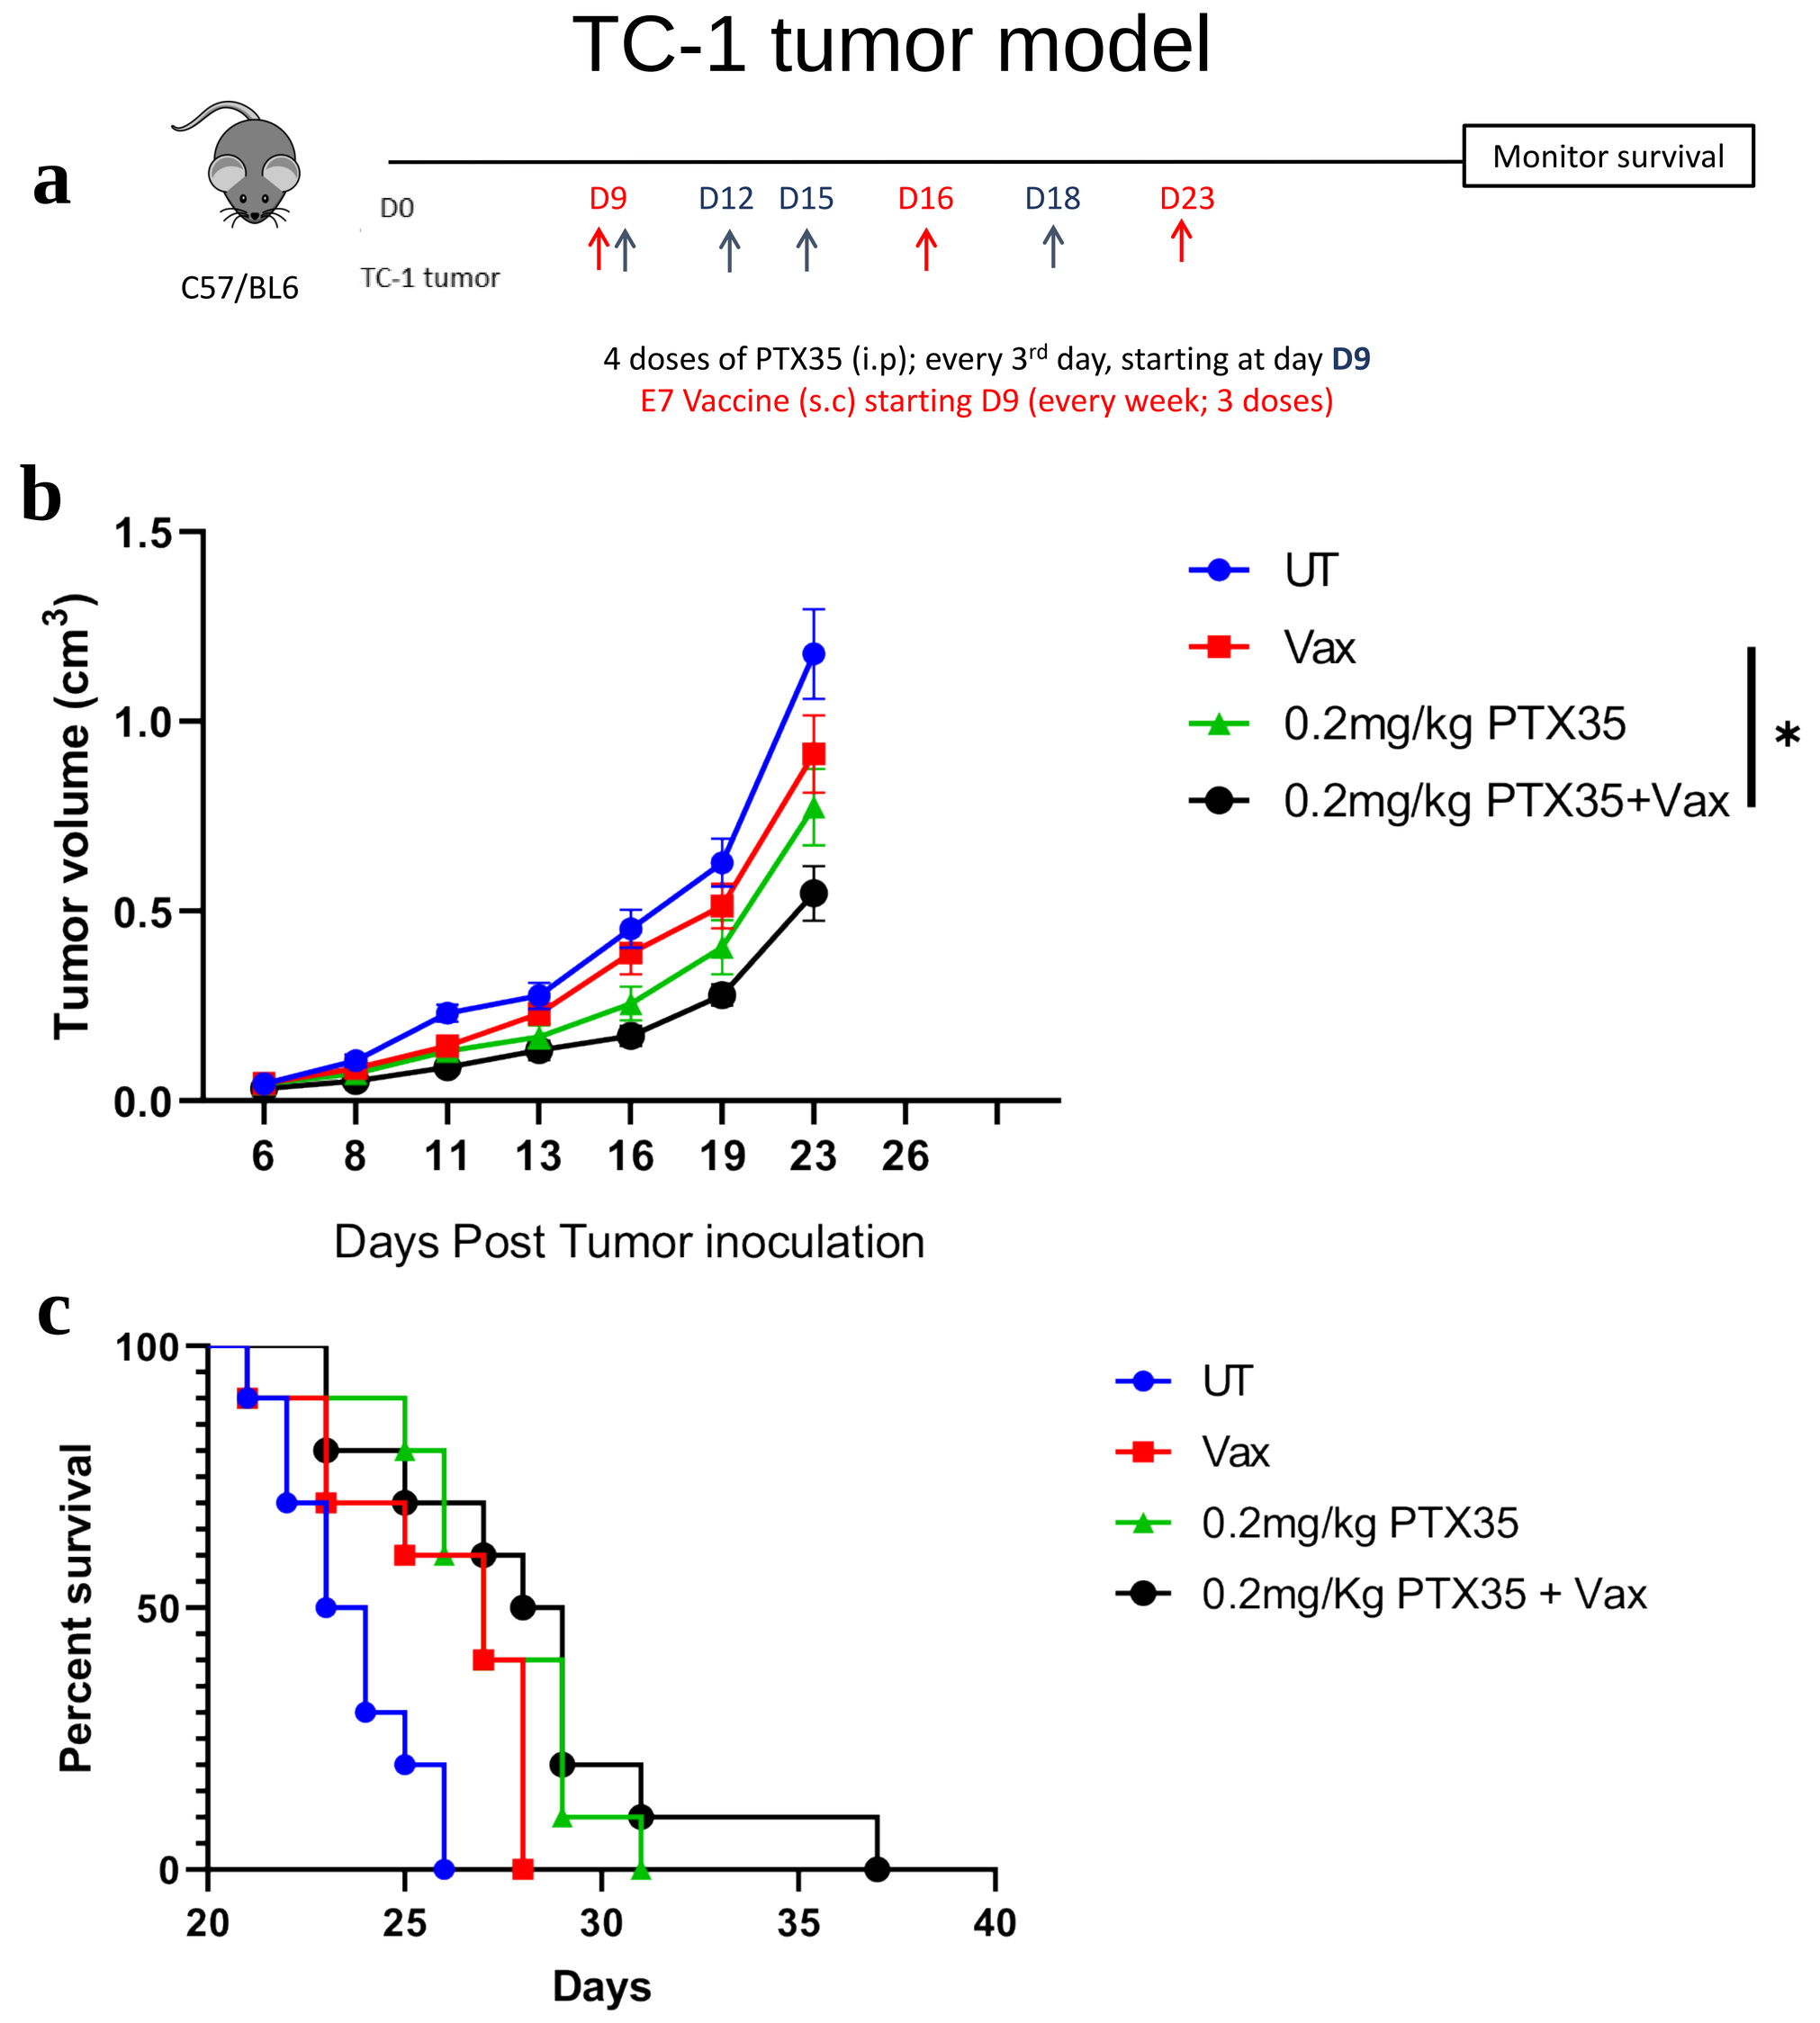

a
C57/BL6
D0
TC-1 tumor
D9 D12 D15 D16 D18 D23
Monitor survival
4 doses of PTX35 (i.p); every 3rd day, starting at day D9
E7 Vaccine (s.c) starting D9 (every week; 3 doses)
b
Tumor volume (cm3)
Days Post Tumor inoculation
UT
Vax
0.2mg/kg PTX35
0.2mg/kg PTX35+Vax
*
c
Percent survival
Days
UT
Vax
0.2mg/kg PTX35
0.2mg/Kg PTX35 + Vax

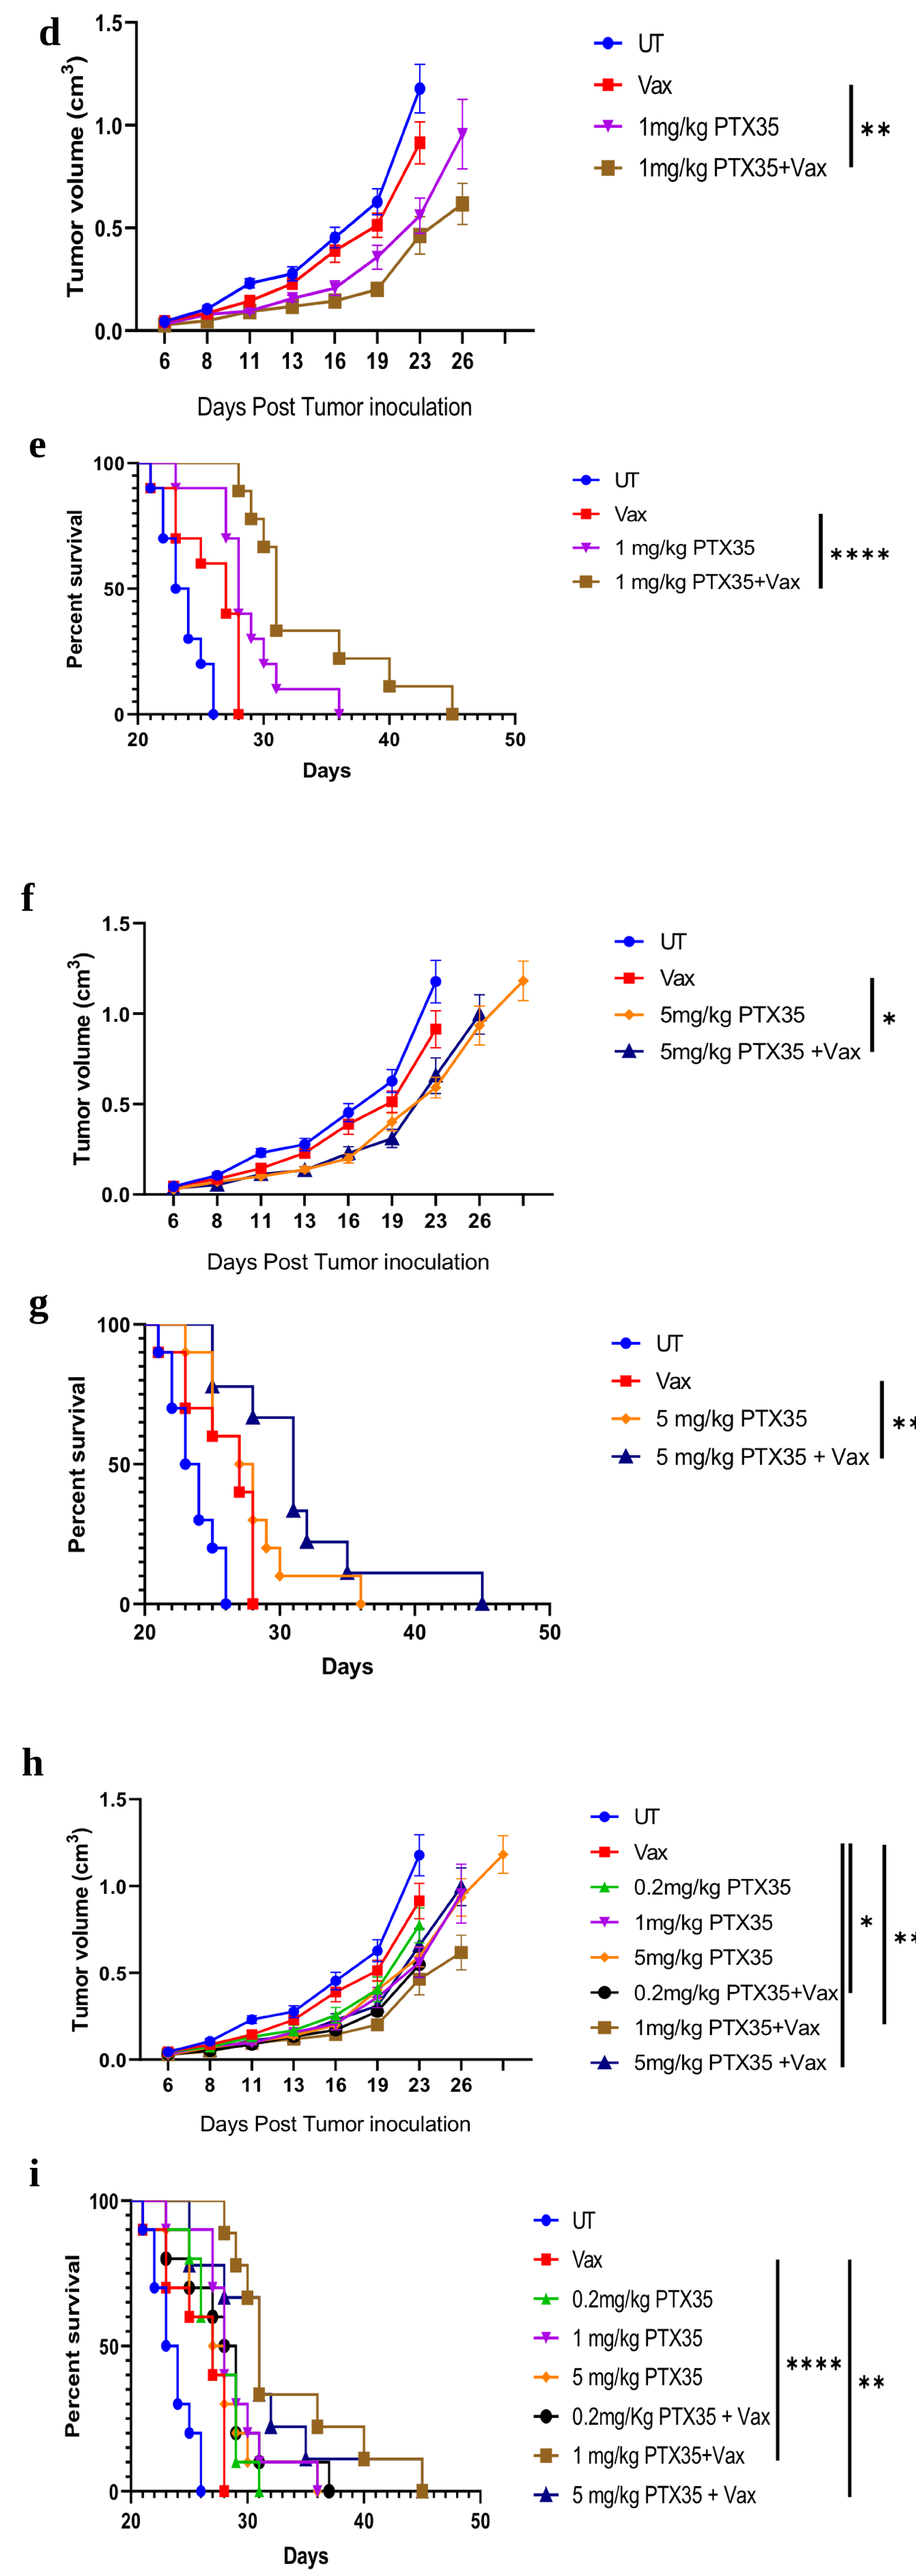

d
Tumor volume (cm3)
Days Post Tumor inoculation
UT
Vax
1mg/kg PTX35
1mg/kg PTX35+Vax
**
e
Percent survival
Days
UT
Vax
1 mg/kg PTX35
1 mg/kg PTX35+Vax

f
Tumor volume (cm3)
Days Post Tumor inoculation
UT
Vax
5mg/kg PTX35
5mg/kg PTX35 +Vax
*
g
Percent survival
Days
UT
Vax
5 mg/kg PTX35
5 mg/kg PTX35 + Vax
**
h
Tumor volume (cm3)
Days Post Tumor inoculation
UT
Vax
0.2mg/kg PTX35
1mg/kg PTX35
5mg/kg PTX35
0.2mg/kg PTX35+Vax
1mg/kg PTX35+Vax
5mg/kg PTX35 +Vax
*
**
i
Percent survival
Days
UT
Vax
0.2mg/kg PTX35
1 mg/kg PTX35
5 mg/kg PTX35
0.2mg/Kg PTX35 + Vax
1 mg/kg PTX35+Vax
5 mg/kg PTX35 + Vax

**

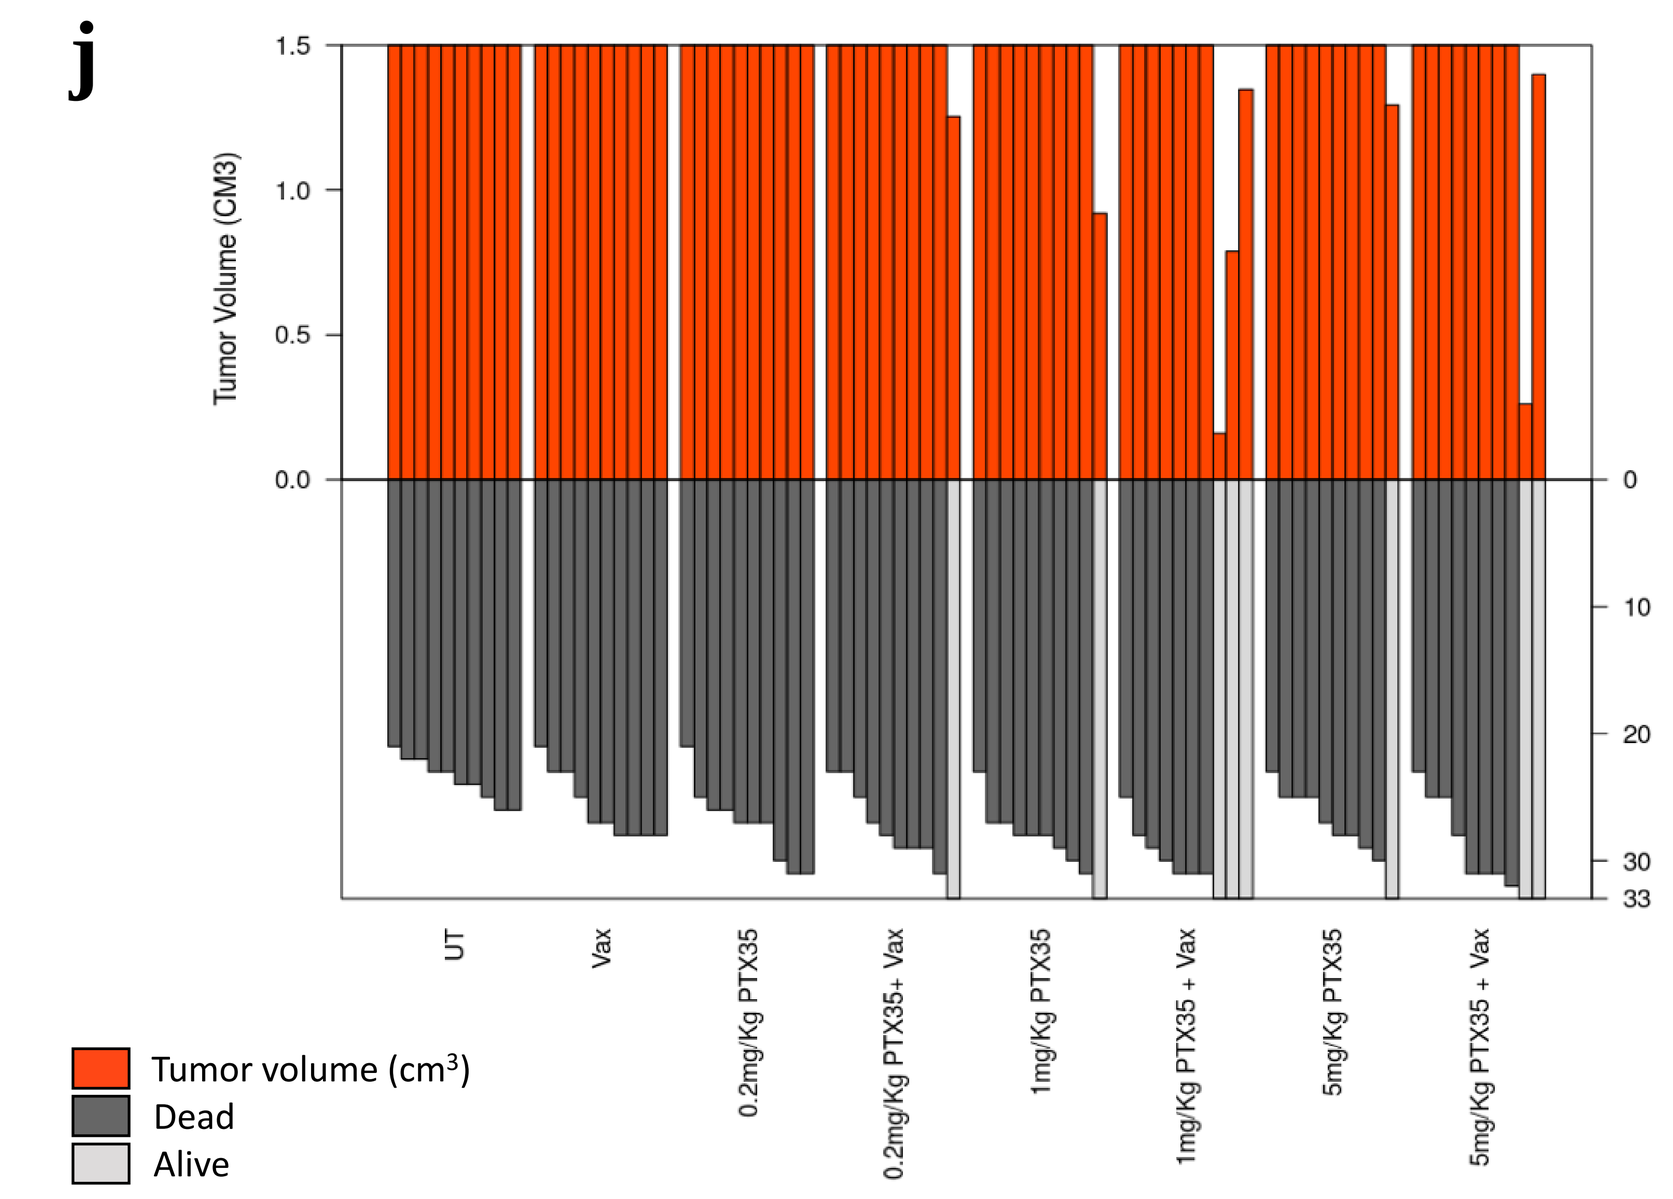

j
Tumor Volume (CM3)
Survival (Days)
UT
Vax
0.2mg/Kg PTX35
0.2mg/Kg PTX35+ Vax
1mg/Kg PTX35
1mg/Kg PTX35 + Vax
5mg/Kg PTX35
5mg/Kg PTX35 + Vax
Tumor volume (cm3)
Dead
Alive

Fig. 1 (a) Schedule of mice treatment. (b) Tumor growth profiles and (c) survival of mice treated with 0.2 mg/kg mPTX-35 , (d) Tumor growth profiles and (e) survival of mice treated with 1 mg/kg mPTX-35, (f) Tumor growth profiles and (g) survival of mice treated with 5 mg/kg mPTX-35, (h) Tumor growth profiles and (i) survival of mice showing all three mPTX-35 doses, (j) SK plot in TC-1 tumor bearing mice. Data are shown from two independent experiments (N=10 mice / treatment group). Statistical significance was calculated using Student's *t*-test with unequal variances for tumor growth and log-rank (Mantel-Cox) tests for survival curves. Data are represented as mean ± SEM. ($^{*}p < 0.05$; $^{**}p < 0.01$; $^{***}p < 0.001$, $^{****}p < 0.0001$).

Proper T cell priming is essential for optimal anti-tumor effects of TNFRSF25 agonist

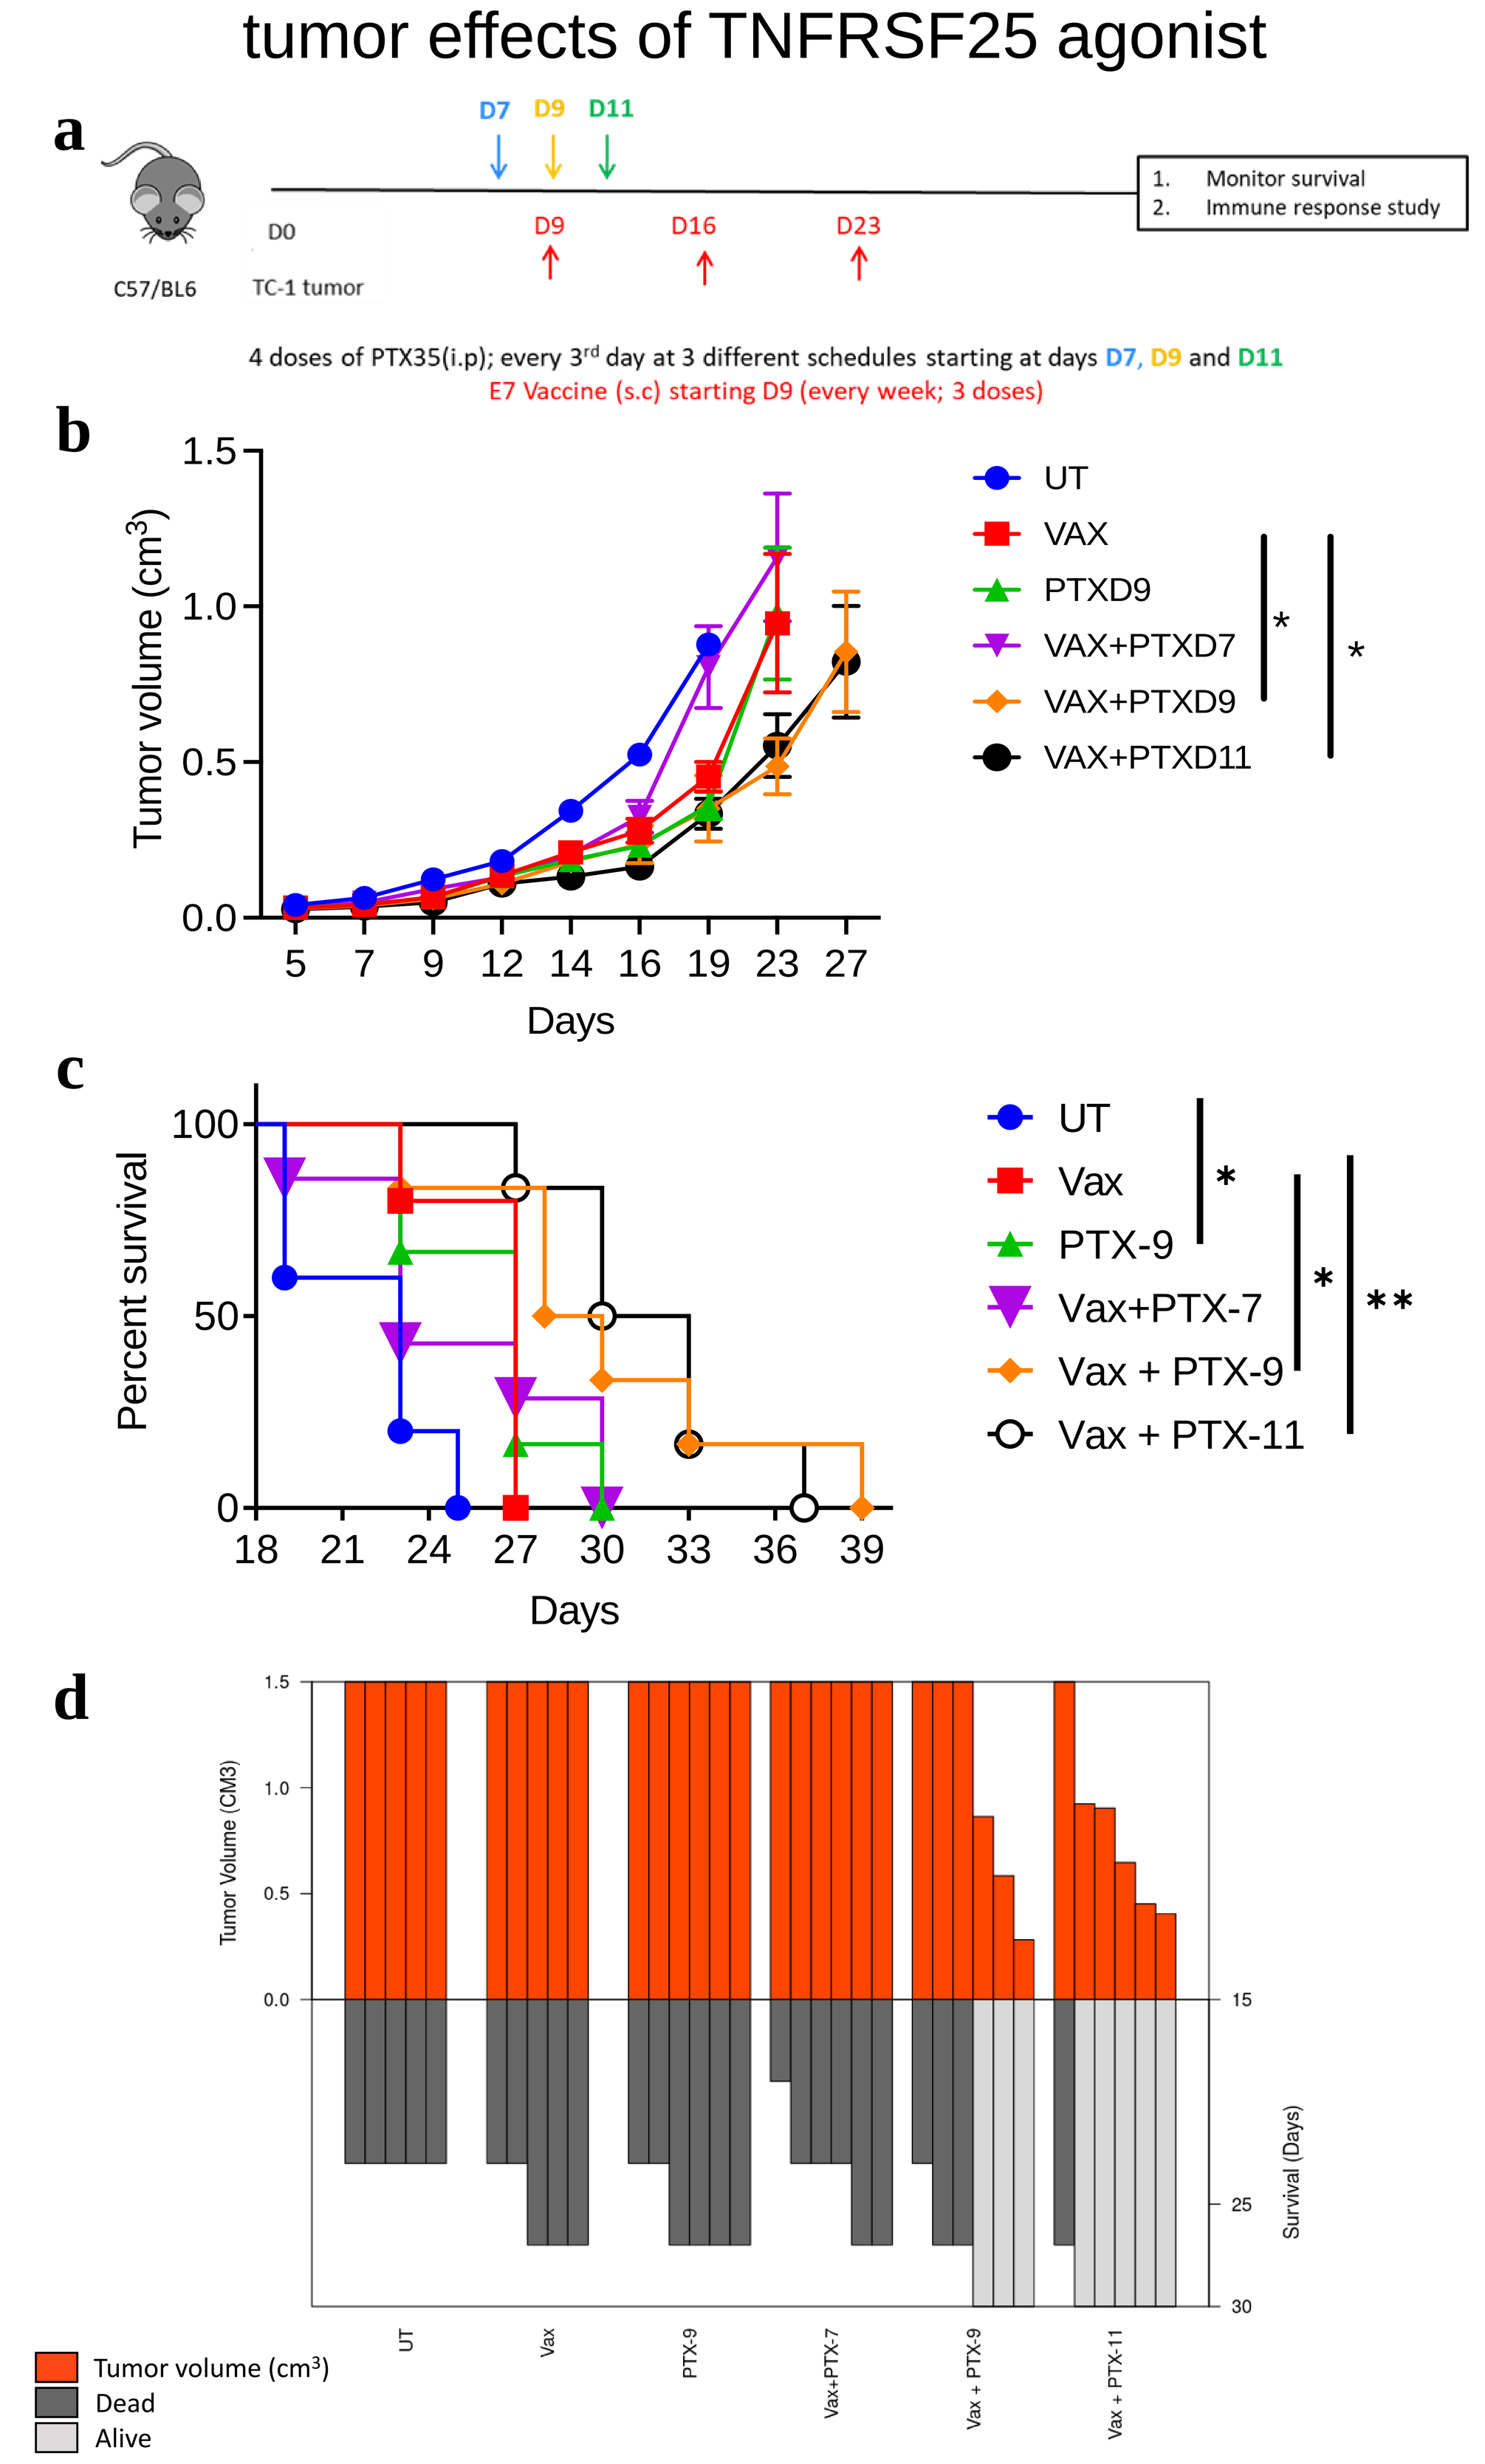

a
C57/BL6
D0
TC-1 tumor
D7 D9 D11
D9 D16 D23
1. Monitor survival
2. Immune response study
4 doses of PTX35(i.p); every 3rd day at 3 different schedules starting at days D7, D9 and D11
E7 Vaccine (s.c) starting D9 (every week; 3 doses)
b
Tumor volume (cm3)
Days
UT
VAX
PTXD9
VAX+PTXD7
VAX+PTXD9
VAX+PTXD11
*
*
c
Percent survival
Days
UT
Vax
PTX-9
Vax+PTX-7
Vax + PTX-9
Vax + PTX-11
*
*
**
d
Tumor Volume (CM3)
Survival (Days)
UT
Vax
PTX-9
Vax+PTX-7
Vax + PTX-9
Vax + PTX-11
Tumor volume (cm3)
Dead
Alive

Fig. 2 (a) Schedule of mice treatment. (b) Tumor growth profiles and (c) mice survival, and (d) SK plot in TC-1 tumor bearing mice after various treatments. Statistical significance was calculated using Student's *t*-test with unequal variances for tumor growth and log-rank (Mantel-Cox) tests for survival curves. Data are represented as mean ± SEM. ($^{*}p < 0.05$; $^{**}p < 0.01$; $^{***}p < 0.001$, $^{****}p < 0.0001$).

TNFRSF25 agonist induces dose-dependent bell-shaped anti-tumor response in the hot CT26 tumor model

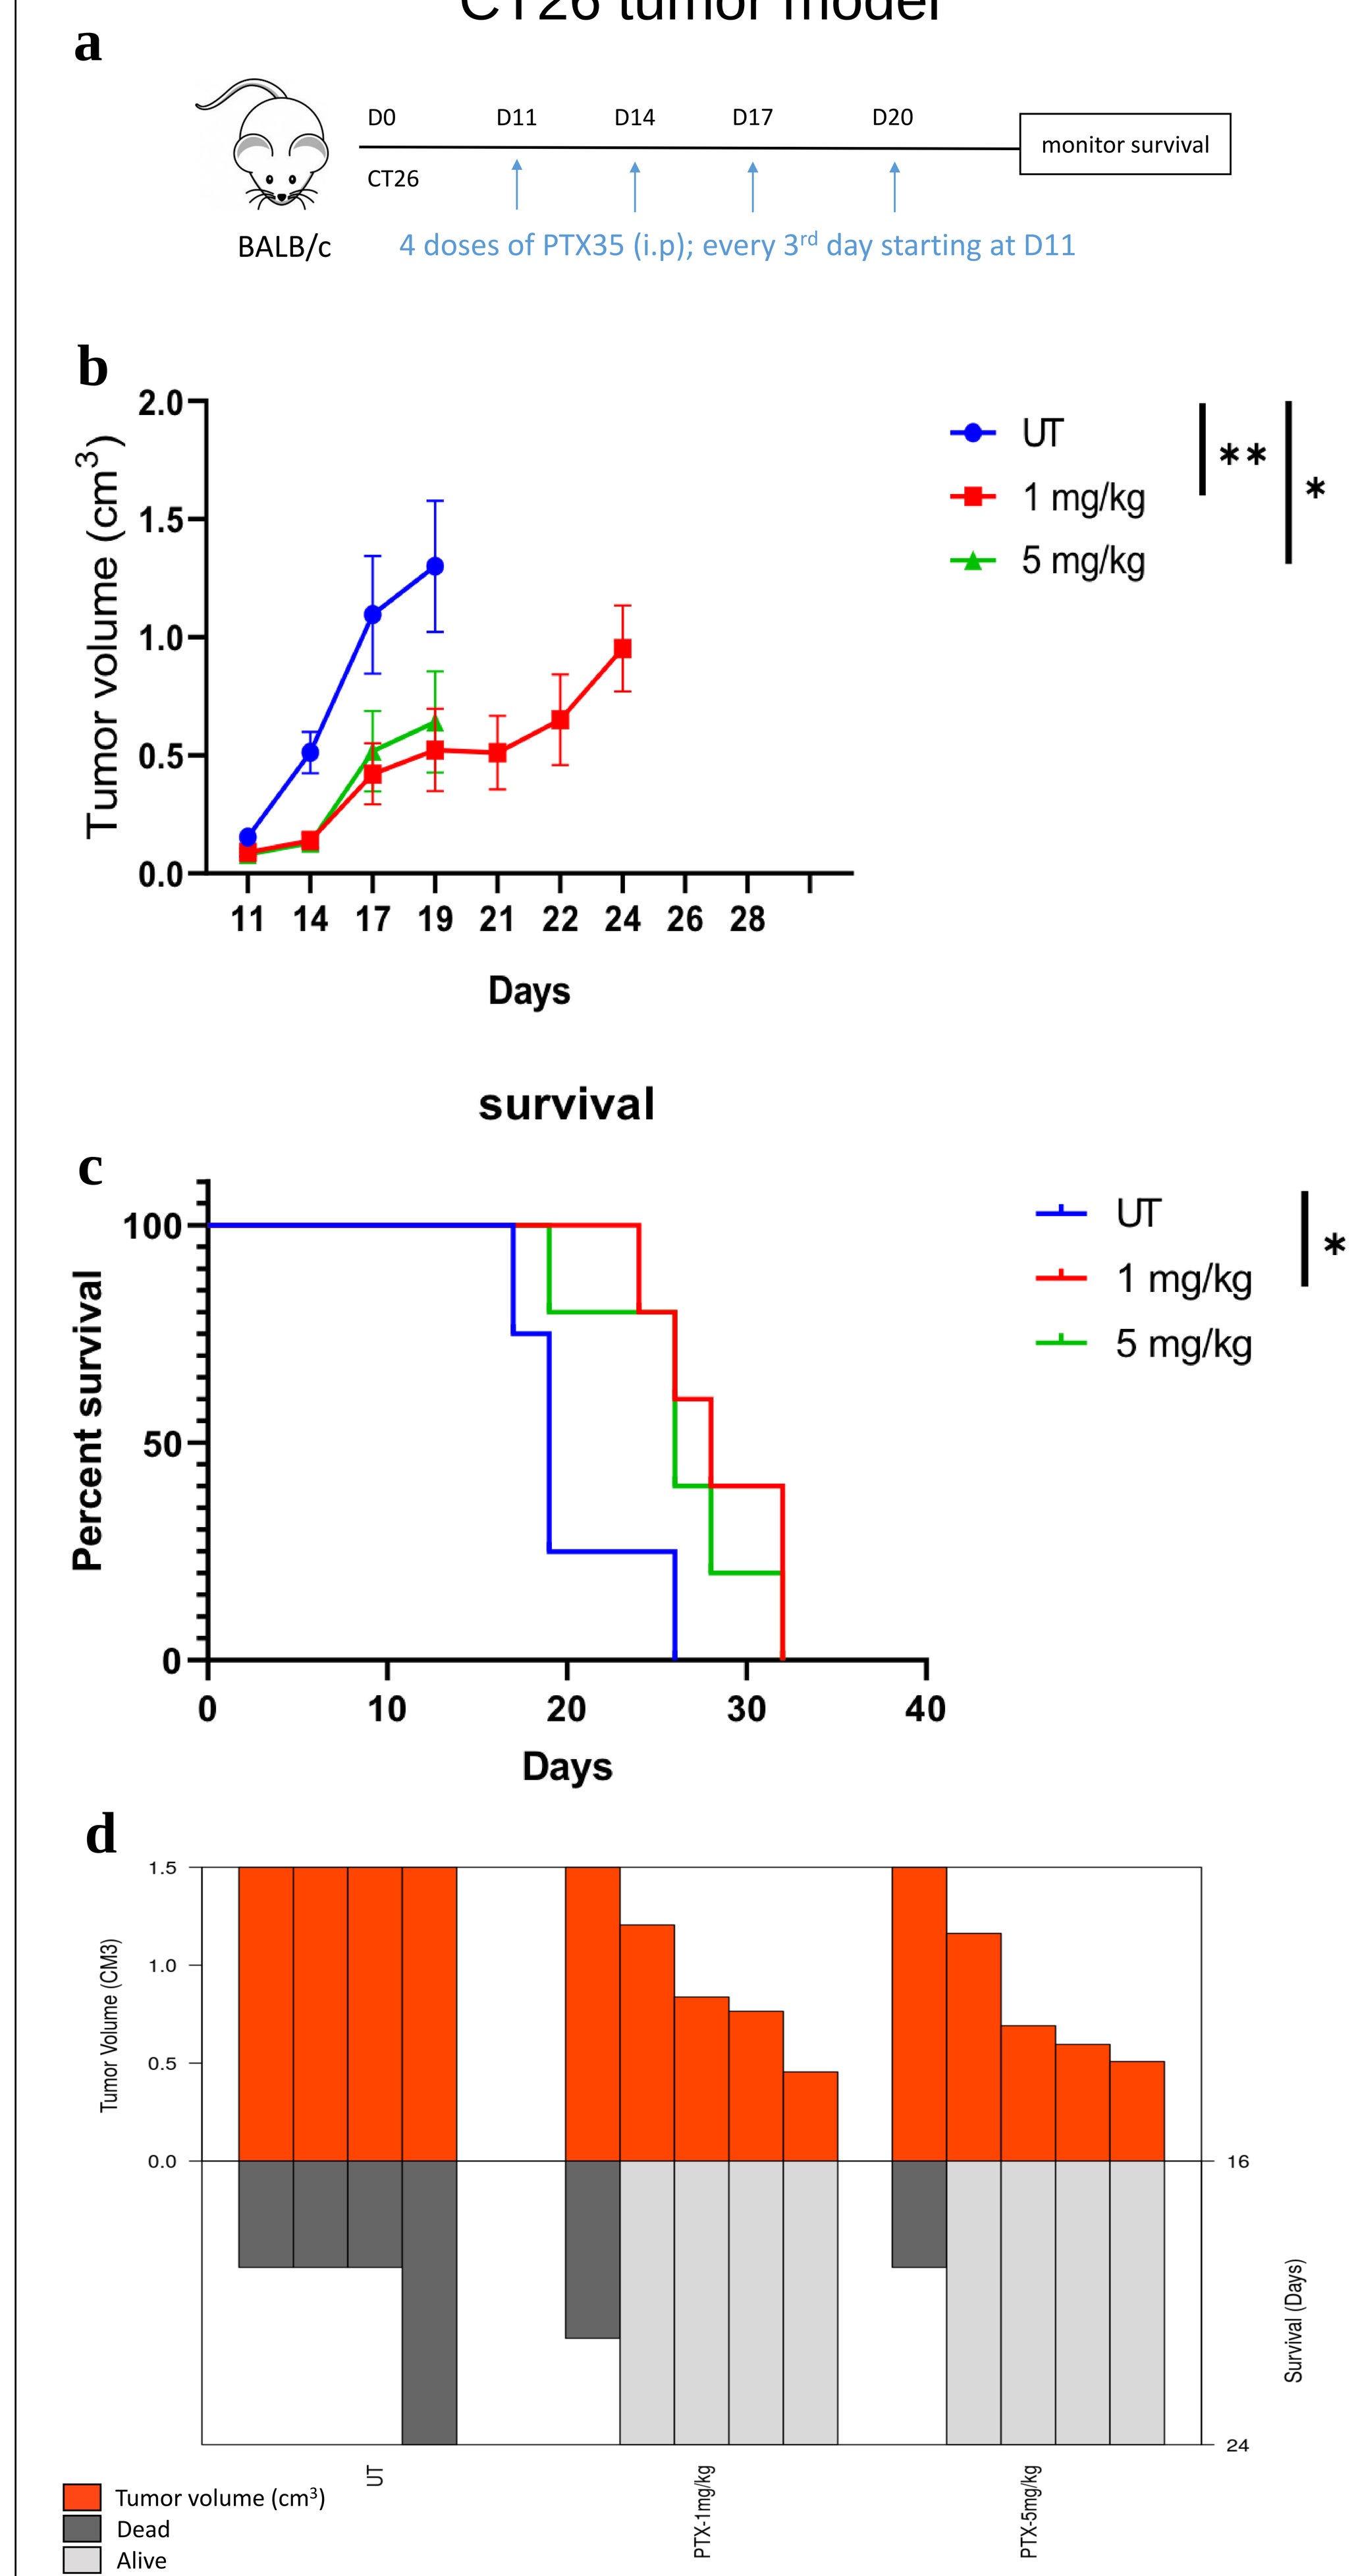

a
BALB/c
D0 D11 D14 D17 D20
CT26
monitor survival
4 doses of PTX35 (i.p); every 3rd day starting at D11
b
Tumor volume (cm3)
Days
UT
1 mg/kg
5 mg/kg
**
*
survival
c
Percent survival
Days
UT
1 mg/kg
5 mg/kg
*
d
Tumor Volume (CM3)
Survival (Days)
UT
PTX-1mg/kg
PTX-5mg/kg
Tumor volume (cm3)
Dead
Alive

Fig. 3 (a) Schedule of mice treatment. (b) Tumor growth profiles and (c) mice survival, and (d) SK plot in CT26 tumor bearing mice after various treatments. Statistical significance was calculated using Student's *t*-test with unequal variances for tumor growth and log-rank (Mantel-Cox) tests for survival curves. Data are represented as mean ± SEM. ($^{*}p < 0.05$; $^{**}p < 0.01$; $^{***}p < 0.001$, $^{****}p < 0.0001$).

Summary and conclusion

Anti-TNFRSF25 agonist antibody demonstrates high anti-tumor efficacy in tumors that are infiltrated with T cells.

Optimal priming is important to limit primary resistance mechanisms and drive an efficacious anti-tumor immune response *via* the TNFRSF25 axis.