DPX™ platform for Drug Delivery: Evaluation of lymph node-targeted delivery of immunomodulatory agents in DPX

ABSTRACT# 6476

Heather Torrey [1], Ava Vila-Leahey [1], Sam Cen [1], Danielle MacKay [1], Alecia MacKay [1], Leanne Lucas [1], Brittany Myers [1], Arthvan Sharma [1], Douglas Vieira [1], Andrea Penwell [1], Rajkannan Rajagopalan [1], and Marianne Stanford [1,2]
[1] IMV, Nova Scotia, Canada; [2] Dalhousie University, Nova Scotia, Canada.

ABSTRACT

Small molecule drugs and biological macromolecules are currently being investigated as therapies for various types of disorders such as cancer and infectious disease. Generally, these agents are systemically administered. However, many immunomodulatory agents are active primarily on specific immune cells such as dendritic cells or T cells. The purpose of this study was to evaluate the efficacy of the novel delivery platform, DPX, for targeted delivery of immunomodulatory agents to the lymph node. As the lymph node is the primary site for priming and activation of immune cells, this approach may improve delivery over systemic administration resulting in increased drug efficacy, the use of less drug, and reduced off-target toxicity.

To evaluate the efficacy of the DPX platform for lymph node-targeted drug delivery, we used the C57Bl/6 murine model. DPX formulation of small molecule dyes allowed us to visualize the kinetics of lymph node-specific delivery by visual assessments and by flow cytometry.

To test impacts on tumor directed immune responses, the HPV16 E7 expressing C3 tumor model was used. Novel formulations were tested in conjunction with DPX-FP (a C3 tumor-specific antigen HPV16 E749-57 conjugated to a T helper epitope). DPX-mediated delivery of low doses of cyclophosphamide (CPA) were delivered subcutaneously as a single injection. Using DPX to deliver CPA resulted in a significant reduction in the number of lymph node cells after a single administration, showing equivalent therapeutic benefit was achieved with significantly less active agent.

We also evaluated DPX delivery of the checkpoint inhibitor anti-CTLA-4. We found that administration in DPX was equivalent to systemic administration by intraperitoneal injection in terms of overall survival of tumor-bearing mice and in the proportion of CD3+ T cells bound by anti-CTLA-4, with fewer administrations of antibody. This demonstrates maintenance of biological functionality of macromolecules in the DPX platform.

In conclusion, DPX holds promise in terms of lymph node-targeted delivery of functional agents. This delivery is facilitated by a sustained uptake from the site of injection and can offer reduced dosing and off-target toxicity as compared to systemic delivery. Many approaches to targeted therapies have failed to provide a therapeutic index commensurate with their promise through systemic delivery, with a challenge in achieving selectivity for the target cell type. Here, we show the DPX drug delivery platform has the potential to overcome this challenge and thereby increase the efficacy of immune and cancer therapies.

METHODS

Novel T Cell Activating Platform

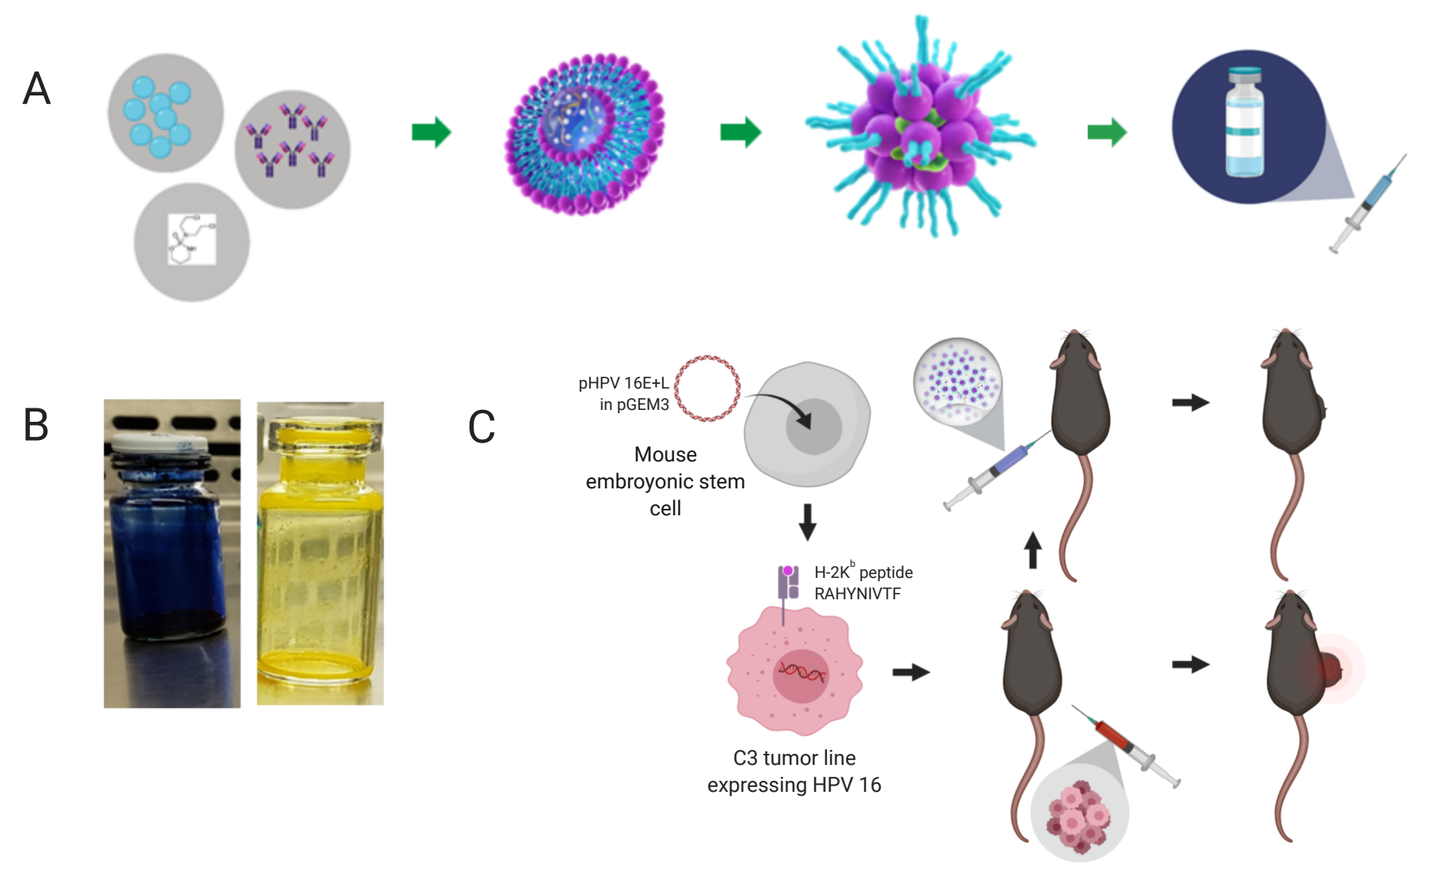
A
B
C
pHPV 16E+L in pGEM3
Mouse embryonic stem cell
H-2K^b peptide RAHYNIVTF
C3 tumor line expressing HPV 16

Figure 1: DPX-forumulated drug delivery and efficacy on tumour control. DPX-formulation of drugs (dyes, biologics, immunomodulatory agents) are prepared in lipid nanoparticles for freeze-drying and distribution (A). Representative dyes formulated in DPX enable visualization of small molecule delivery (B). Functional efficacy of DPX-delivery is evaluated in mice implanted with HPV16 E7 expressing C3 tumour model (C).

Distribution

DPX-formulation of small molecule dyes demonstrated targeted delivery compared to systemic aqueous delivery

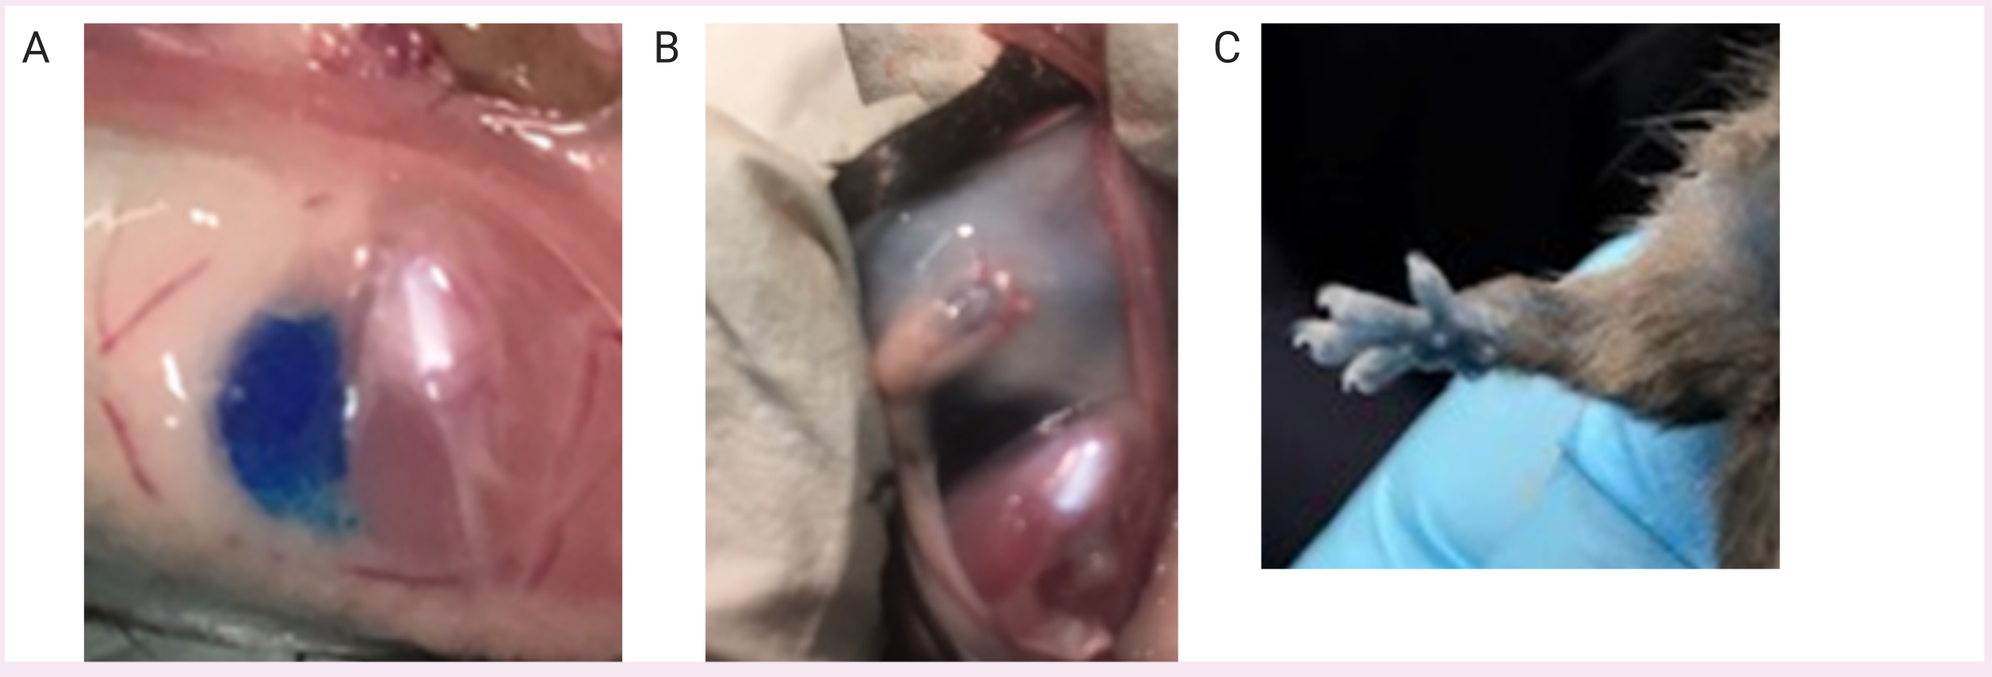
A
B
C

Figure 2. DPX-dyes result in localized versus systemic distribution. DPX-based delivery demonstrates a clear sequestration of the dye in the subcutaneous depo (A). The dye can then be visualized within the regional lymph node within a couple of days (B) which was not seen in untreated mice (image unavailable). Aqueous formulations demonstrate widespread dissemination of the dye as demonstrated by blue dye visualized in the skin (C) while DPX-dye treated mice had normal skin colour (image unavailable).

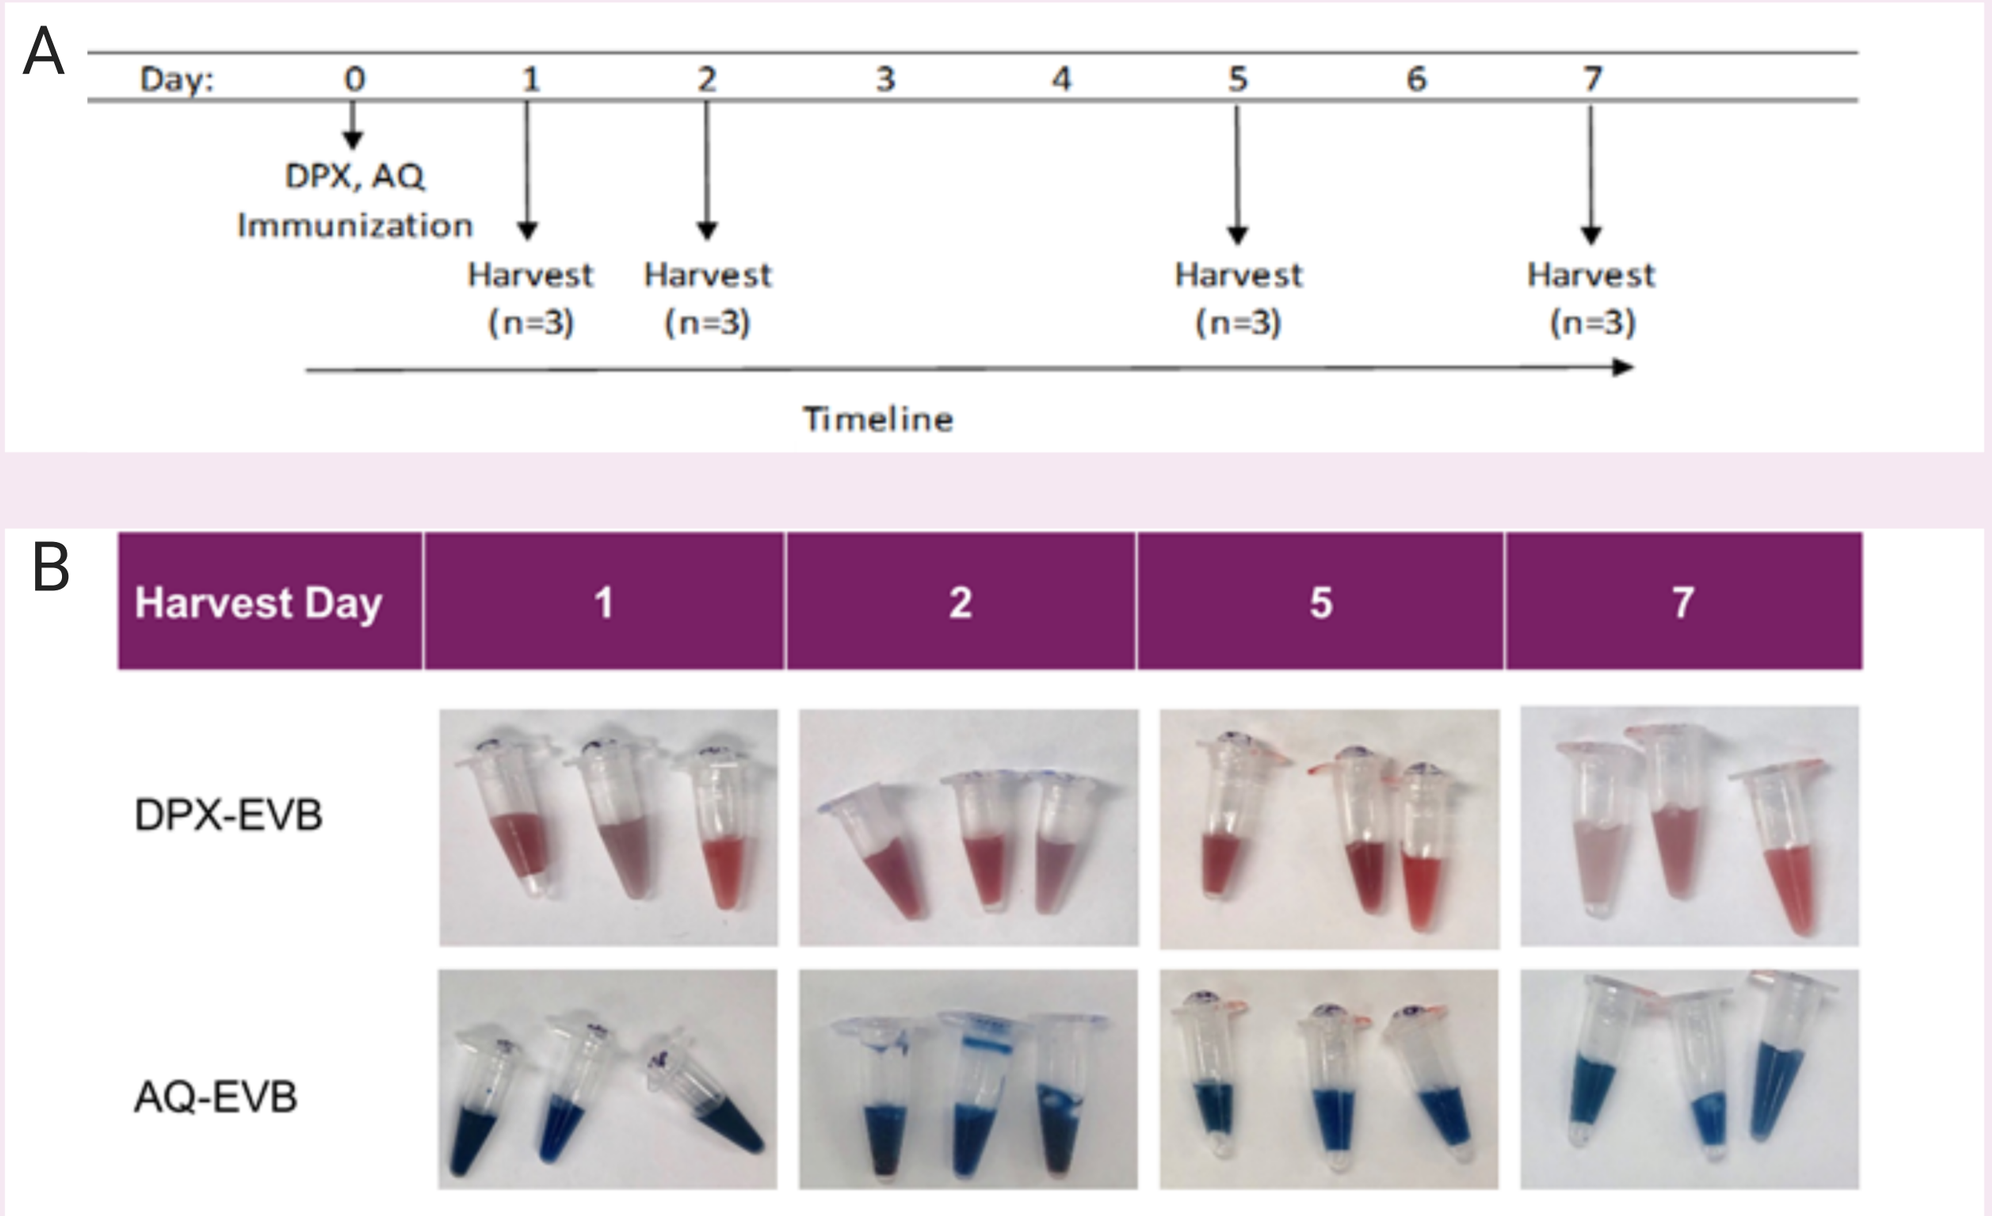
A
Day: 0 1 2 3 4 5 6 7
DPX, AQ Immunization
Harvest (n=3)
Harvest (n=3)
Harvest (n=3)
Harvest (n=3)
Timeline
B
Harvest Day 1 2 5 7
DPX-EVB
AQ-EVB

Figure 3. Visual inspection of serum samples from mice treated with DPX-dye versus Aq-Dye. Study schedule of C57Bl/6 mice (n=3 per time point) injected subcutaneously on study day 0 with 1 mg of Evans Blue (EVB) dye formulated either in DPX or in an aqueous solution (A). Comparison of serum from DPX-EVB formulations with aqueous-EVB formulations demonstrates that with DPX there is minimal to no systemic exposure (B).

Figure 4: Panel 1 - EVB

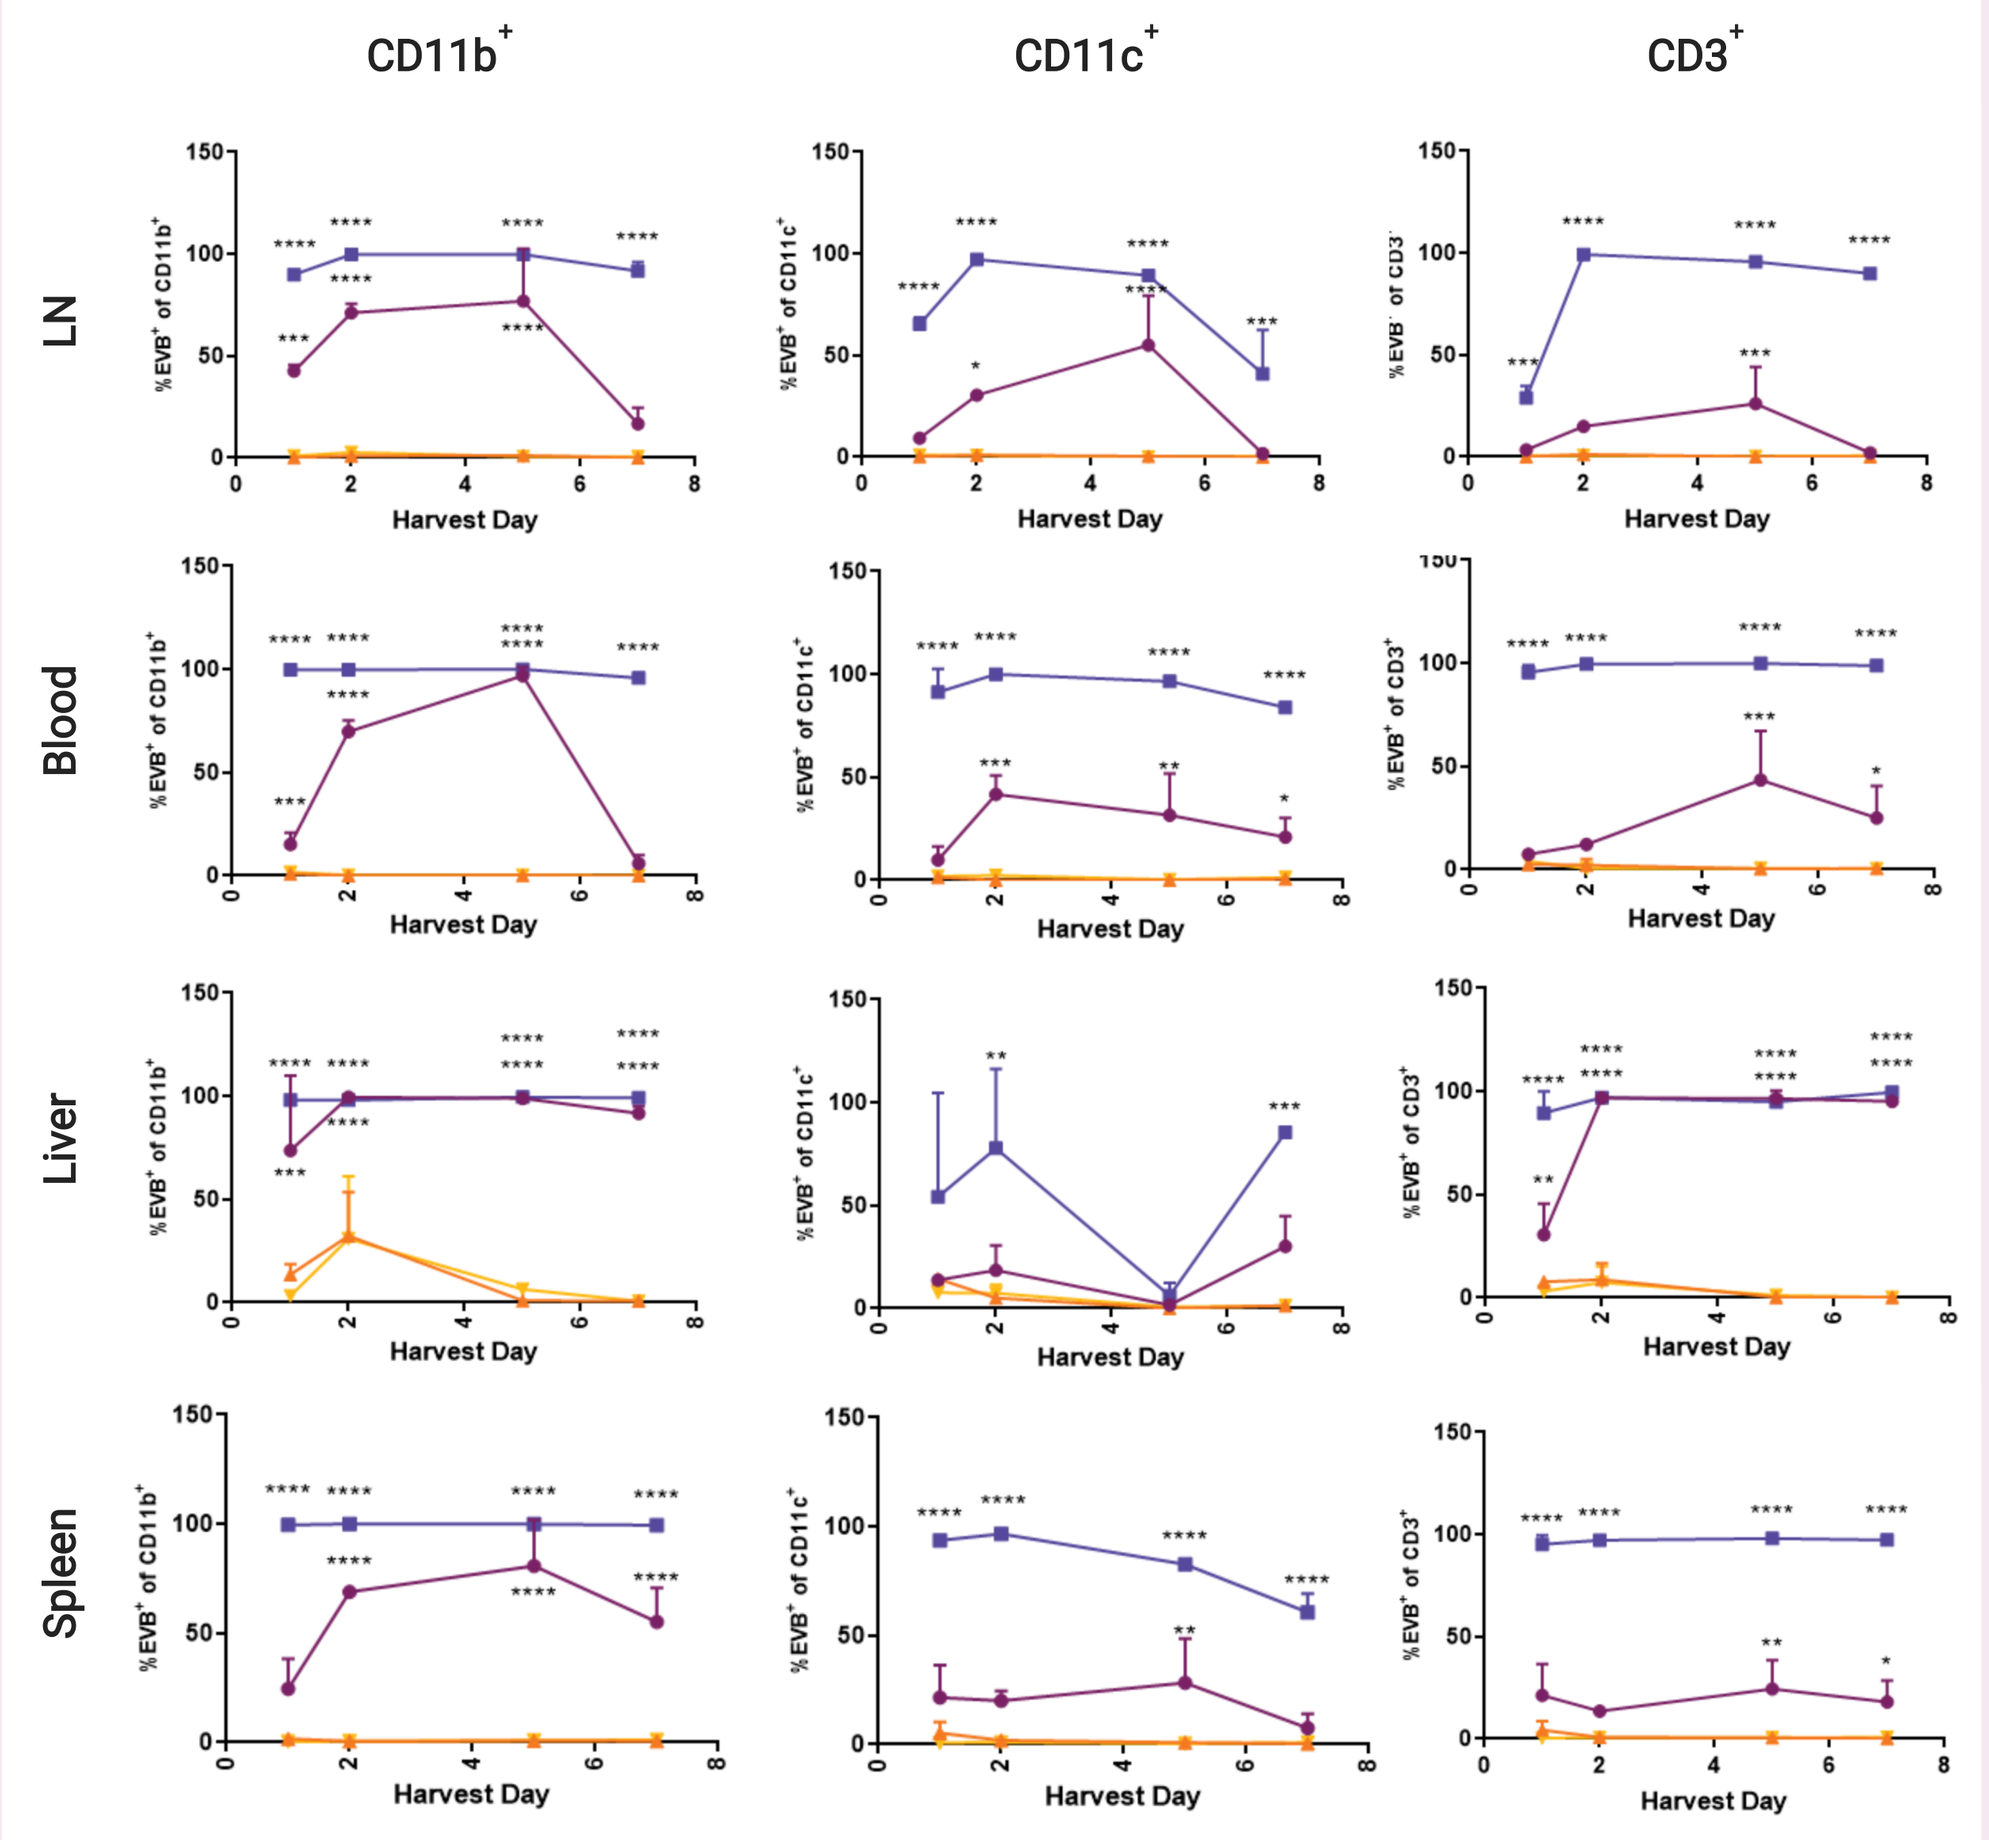
CD11b+
CD11c+
CD3+
LN
Blood
Liver
Spleen
Harvest Day

Figure 4: Panel 2 - AF488

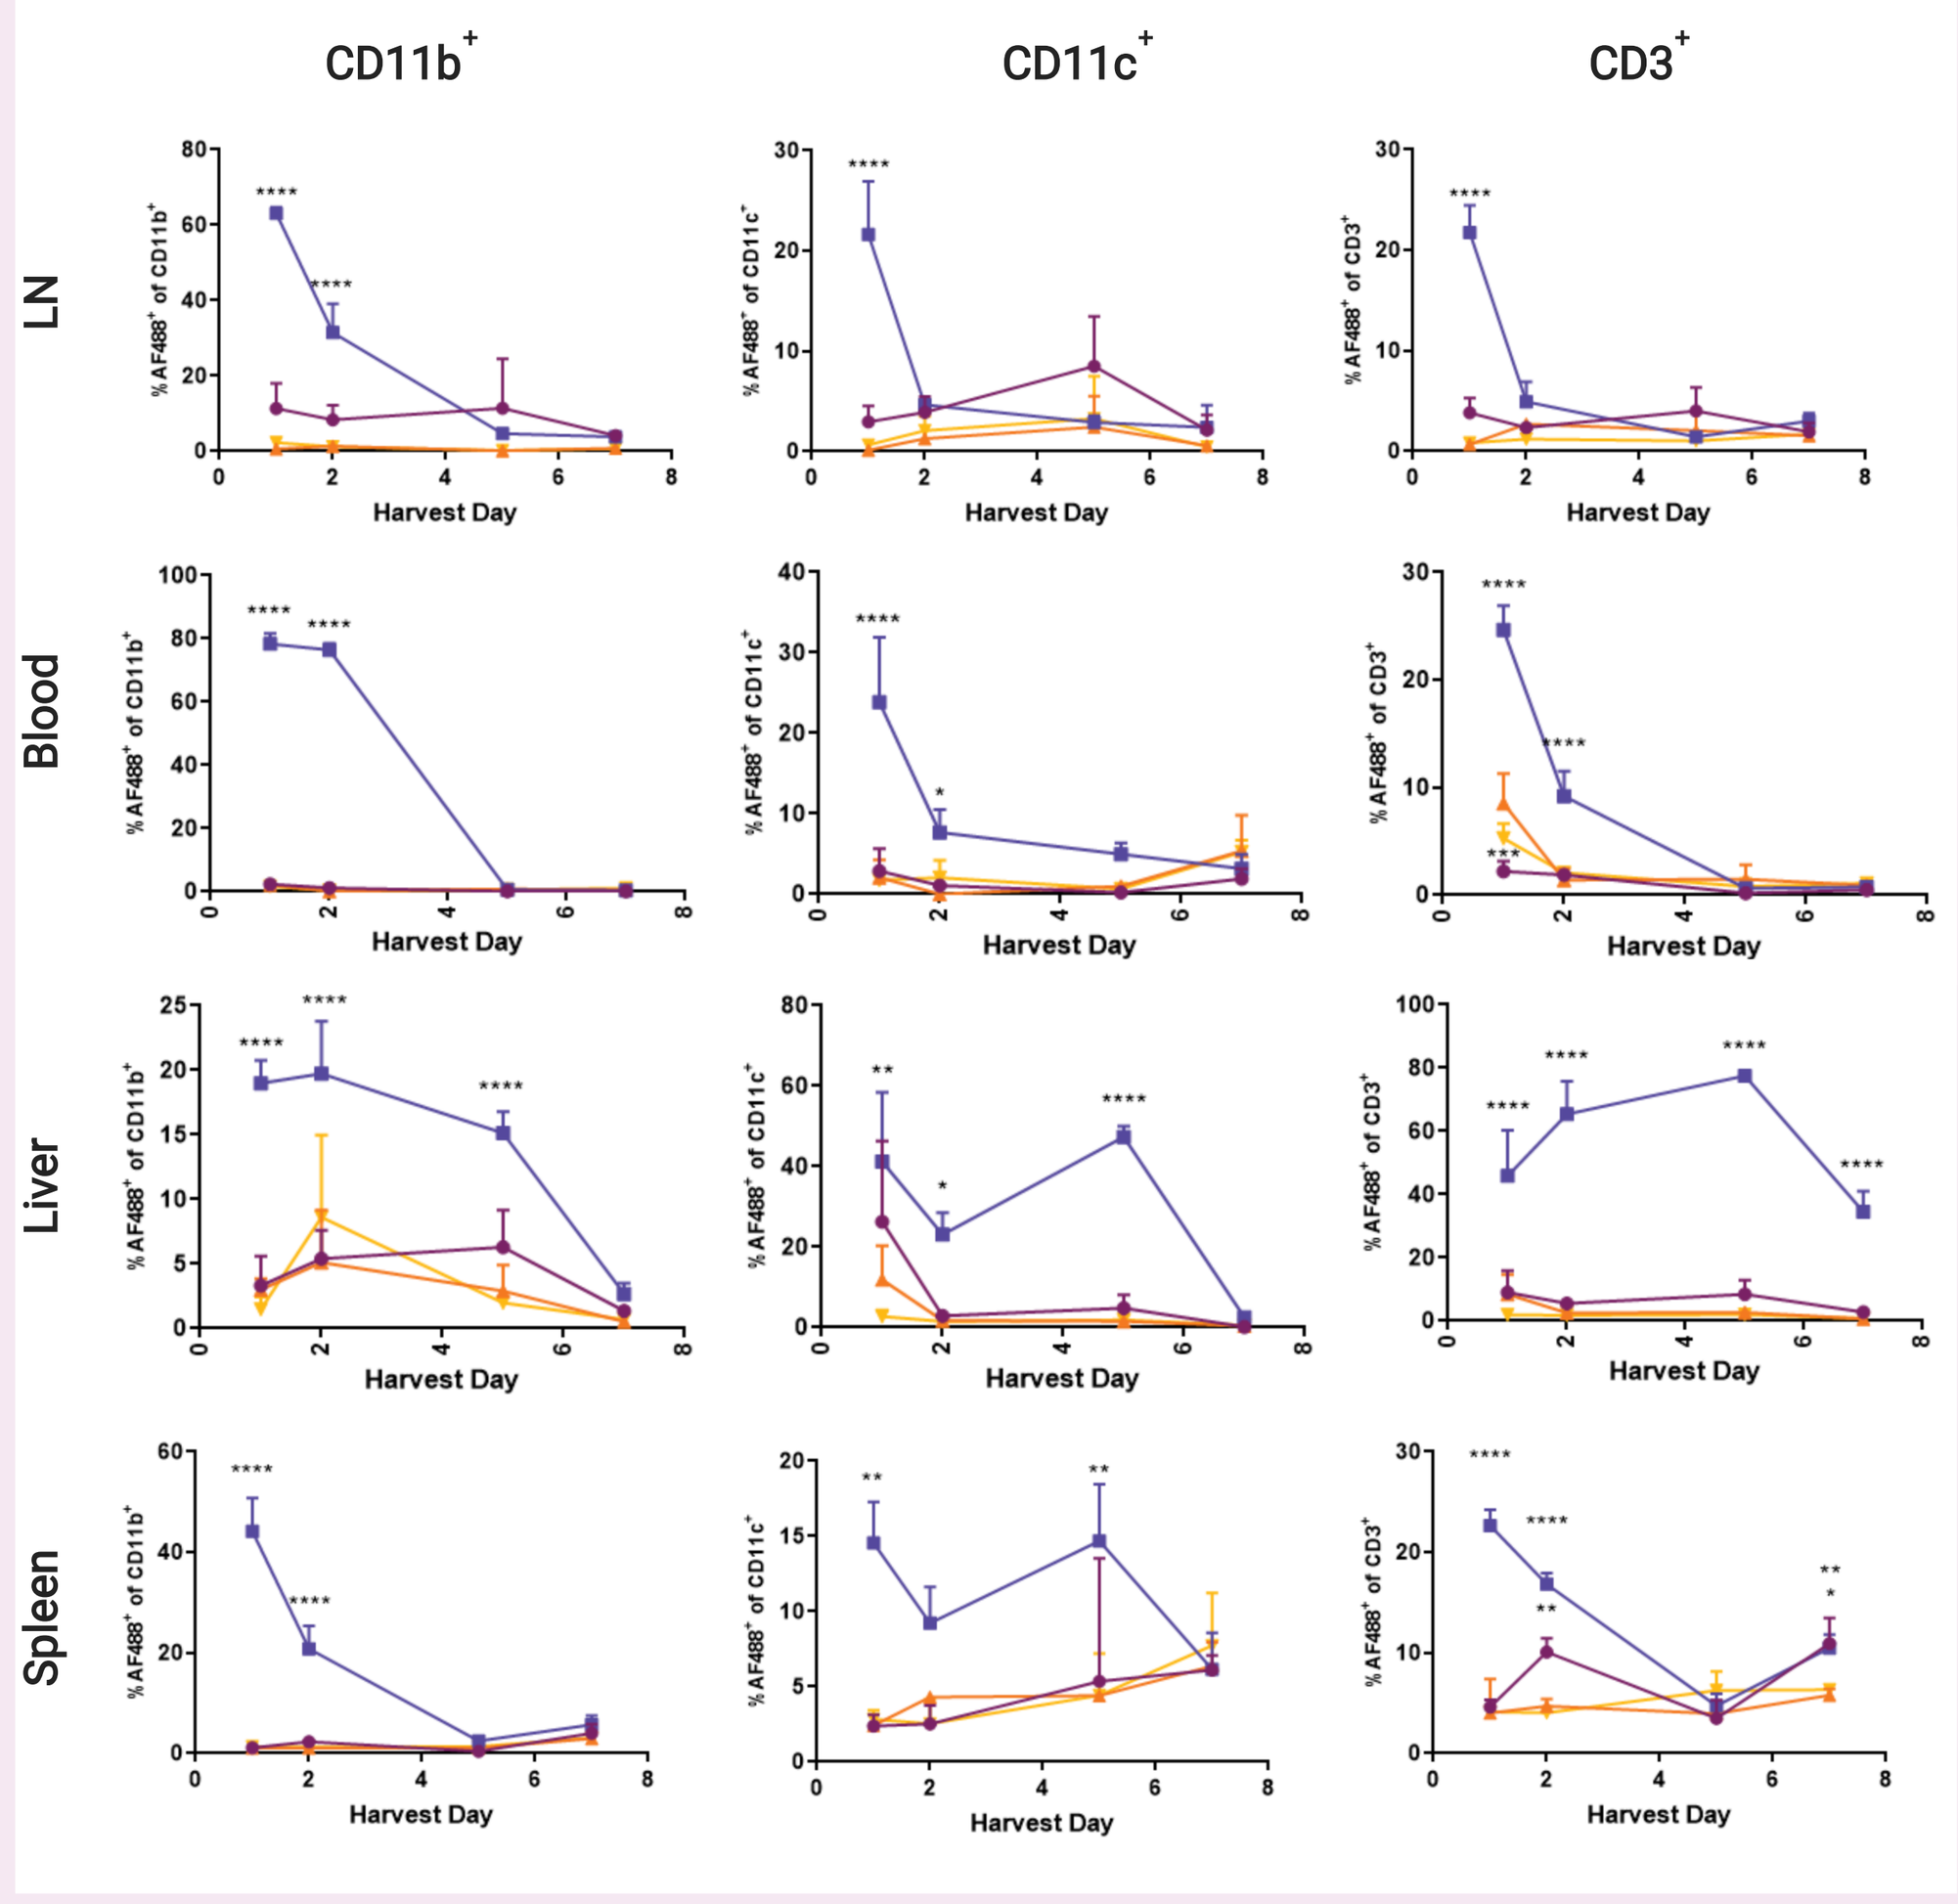
CD11b+
CD11c+
CD3+
LN
Blood
Liver
Spleen
Harvest Day

Fgure 4 Flow cytometric analysis of dye delivery by DPX compared to aqueous formulation. Percentage of various cell types ($CD11b^+$ macrophages A, D, G, J; $CD11c^+$ dendritic cells B, E, H, K; $CD3^+$ T cells C, F, I, L) from tissues (LN A, B, C; Blood D, E, F; Liver G, H, I; Spleen J, K, L) longitudinally (Days 1, 2, 5, and 7) that contained EVB (Panel 1) and AF488 (Panel 2) as assessed by flow cytometry. EVB (MW 960.81 g/mol) is a membrane impermeant dye with high affinity for serum albumin that can penetrate non-viable cells or be actively internalized by phagocytic cells, such as macrophages and dendritic cells. AF488 (MW 773.91 g/mol) is a membrane impermeant fluorescent dye that can be actively internalized by phagocytic cells, such as macrophages and dendritic cells. Experimental groups include DPX-dye (Group 1; purple circles), aqueous-dye (Group 2; blue squares), while control groups were injected subcutaneously with DPX-empty (Group 3; orange upwards triangles) or not injected (Group 4; yellow downwards triangles).

Targeted Delivery

DPX-drug delivery achieved equivalent LN-specific effects with significantly less active agent

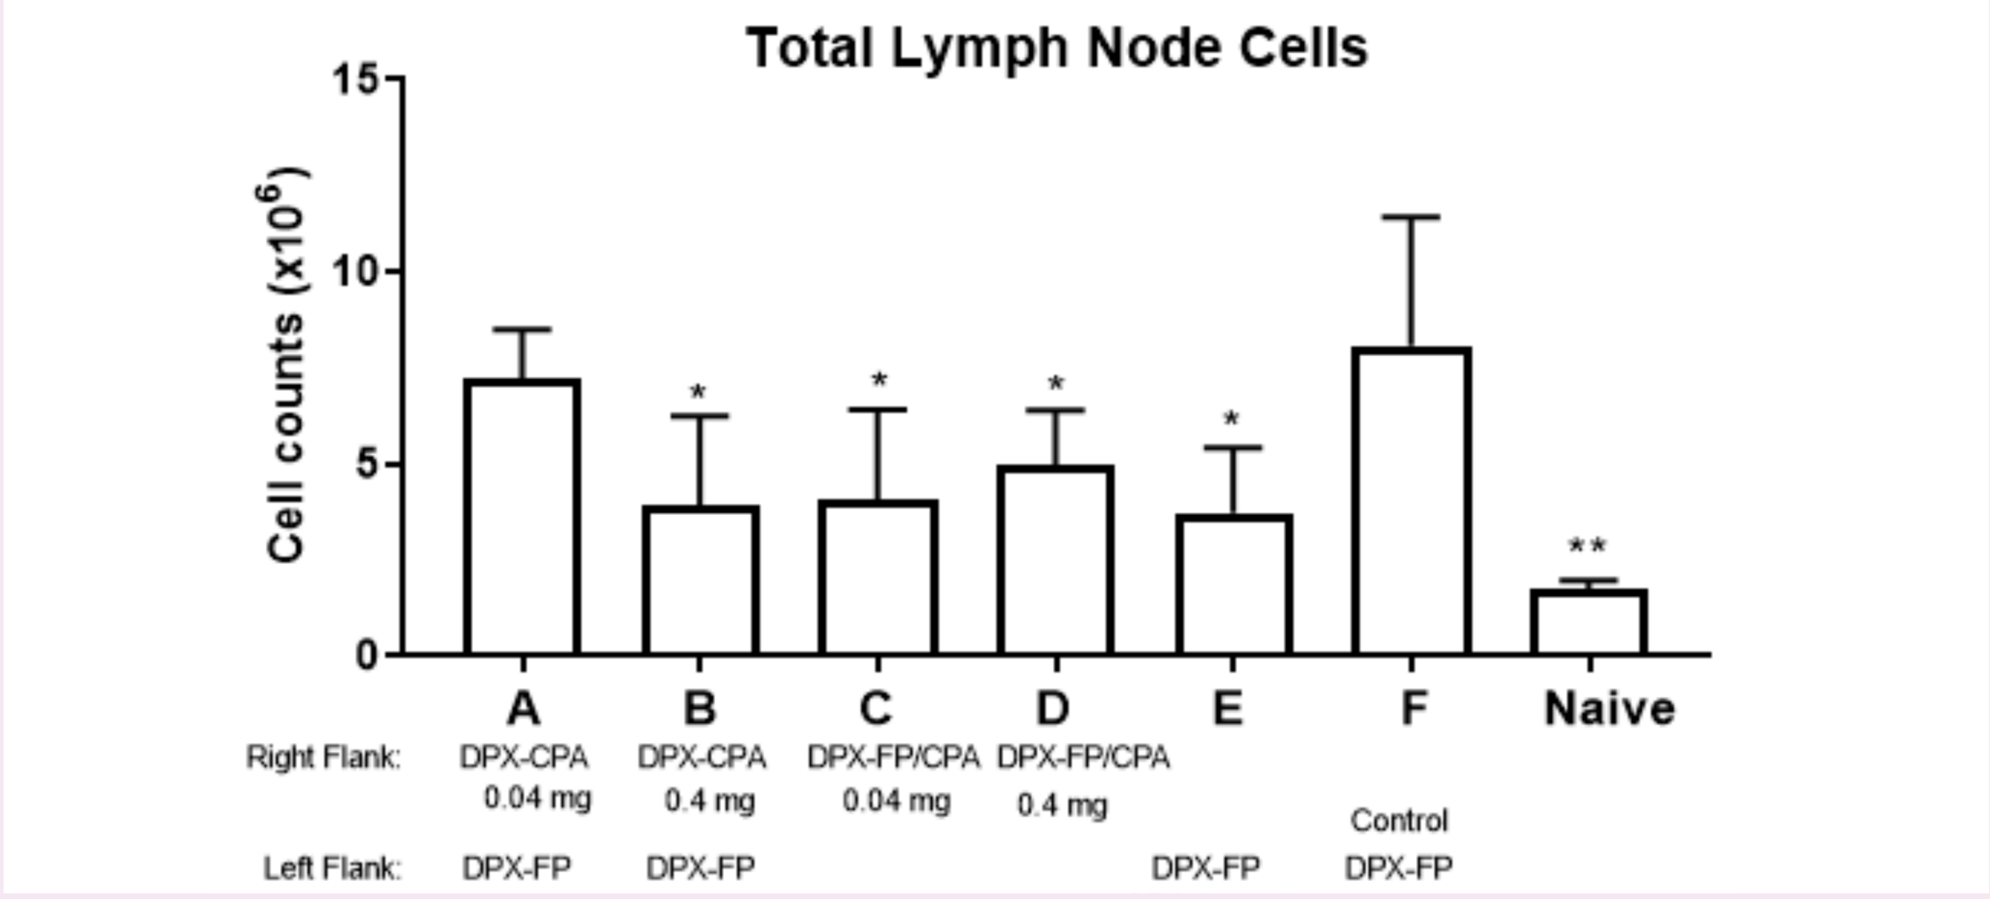
Total Lymph Node Cells
Cell counts (x10^6)
A B C D E F Naive
Right Flank: DPX-CPA 0.04 mg DPX-CPA 0.4 mg DPX-FP/CPA 0.04 mg DPX-FP/CPA 0.4 mg
Control
Left Flank: DPX-FP DPX-FP DPX-FP DPX-FP

Figure 5: Total lymph node cells depleted by DPX-CPA. Low doses of the small molecule CPA was formulated in DPX and 50 µl (0.04 mg A, C; 0.4 mg B, D) was injected in the right flank of mice (n=5 per group) with DPX-FP (left flank A, B, E, F; right flank C, D) and total lymph node cells were counted. Mice in group E were treated for seven days prior to injection with 20 mg/kg/day CPA provided in drinking water, which corresponds to 0.4 mg per day for seven days. All mice were terminated 8 days following injections and inguinal lymph nodes were processed in a single cell suspension and counted. Statistical analysis by unpaired T test control (DPX-FP), one-tailed, **p <0.01, *p < 0.05.

Functionality

DPX formulated with functional biological molecules

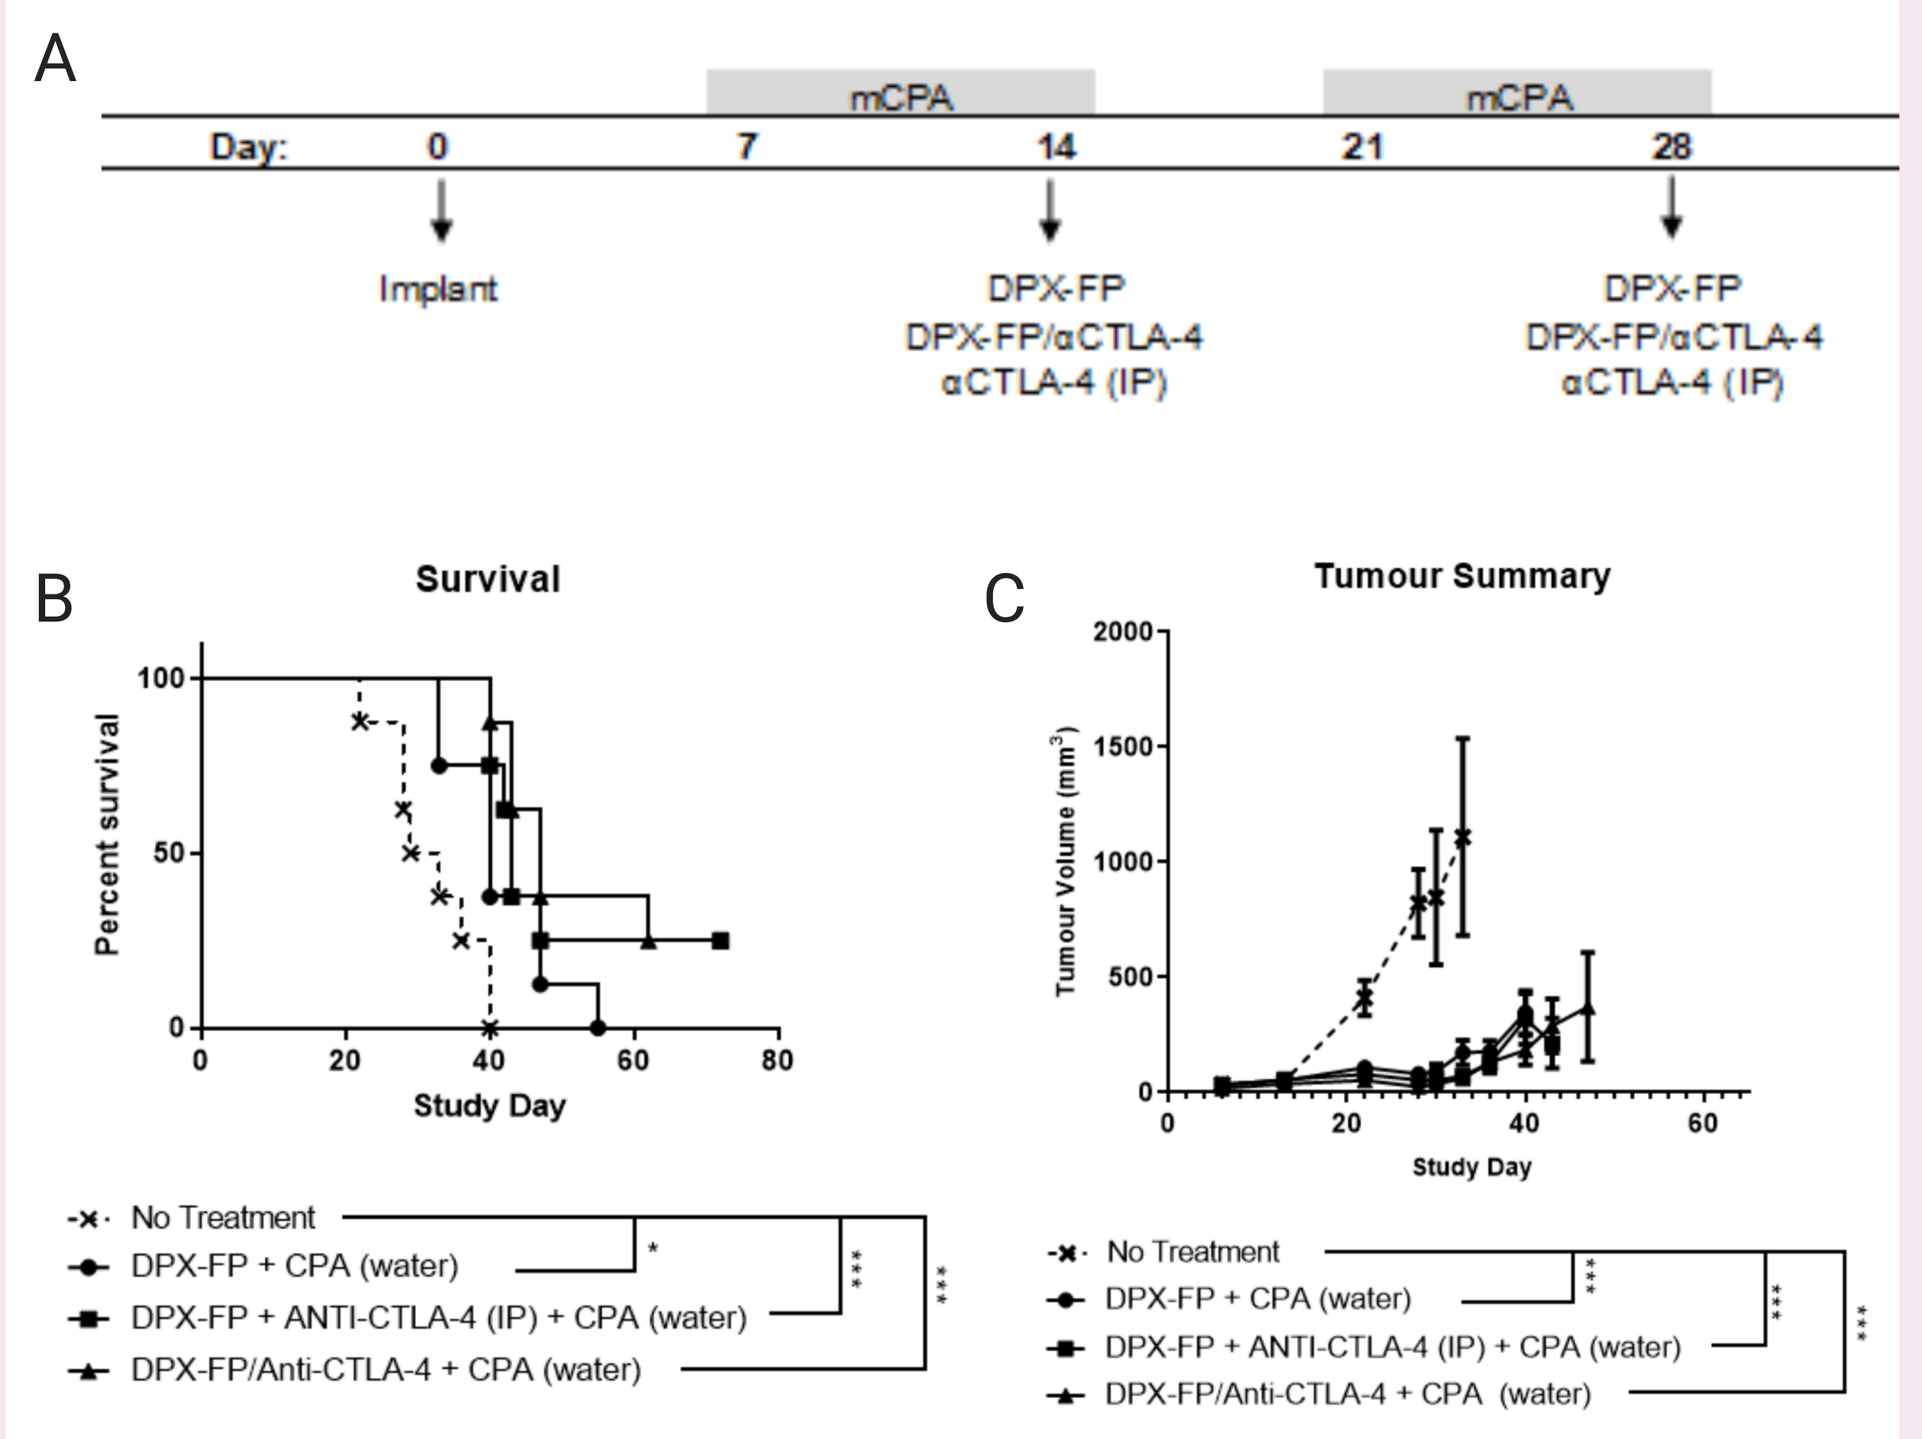
A
Day: 0 7 14 21 28
mCPA mCPA
Implant
DPX-FP
DPX-FP/αCTLA-4
αCTLA-4 (IP)
B
Survival
Percent survival
Study Day
C
Tumour Summary
Tumour Volume (mm^3)
Study Day
No Treatment
DPX-FP + CPA (water)
DPX-FP + ANTI-CTLA-4 (IP) + CPA (water)
DPX-FP/Anti-CTLA-4 + CPA (water)

Figure 6: Mice treated with DPX-FP/anti-CTLA-4 exhibited a significantly improved survival rate and reduction in tumour growth. With the 28-day treatment schedule (A), we find that the survival rate (B) and reduction in tumour growth (C) of mice (n=8) treated with DPX formulation is equivalent to systemic anti-CTLA-4 by IP injection compared to untreated mice. This demonstrates that CPA plus anti-CTLA-4 delivered in DPX effectively restricted tumour growth and improved survival equivalently to administration of CPA plus anti-CTLA-4 through systemic delivery by IP injection.

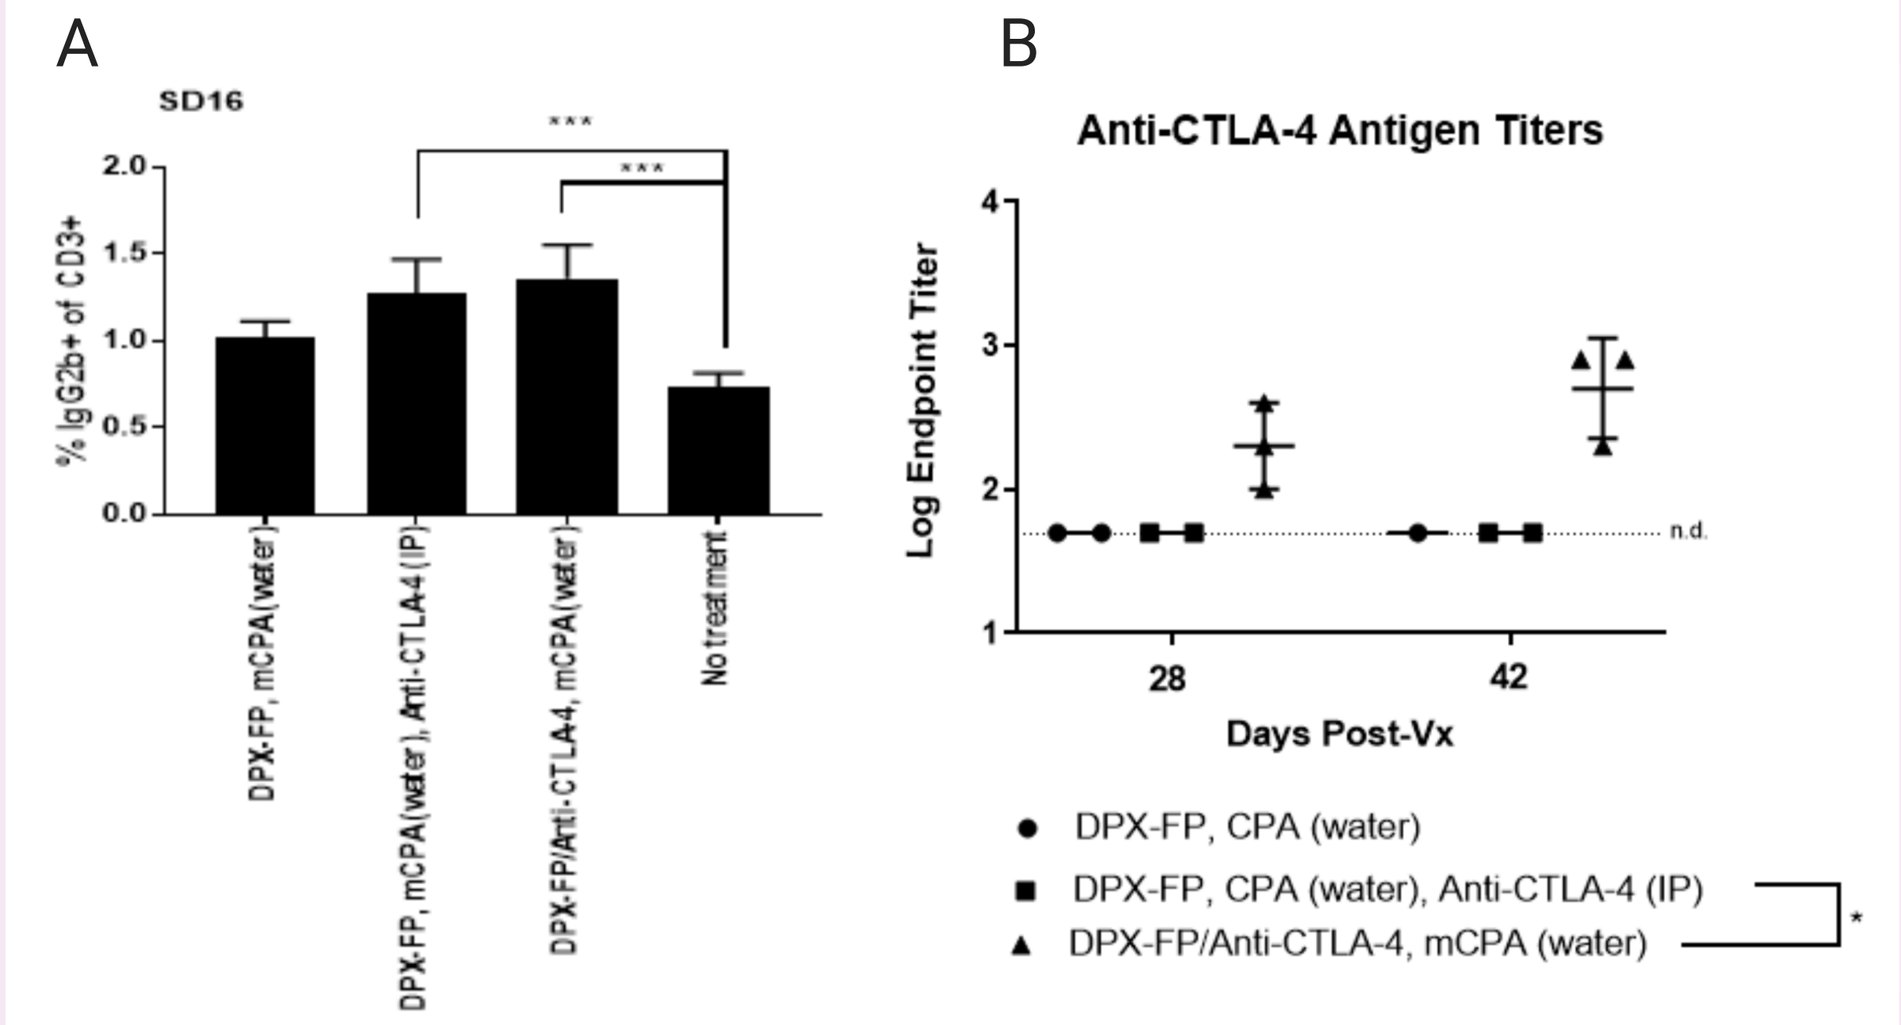
A
SD16
% IgG2b+ of CD3+
DPX-FP, mCPA(water)
DPX-FP, mCPA(water), Anti-CTLA4 (IP)
DPX-FP/Anti-CTLA4, mCPA(water)
No treatment
B
Anti-CTLA-4 Antigen Titers
Log Endpoint Titer
Days Post-Vx
n.d.
DPX-FP, CPA (water)
DPX-FP, CPA (water), Anti-CTLA-4 (IP)
DPX-FP/Anti-CTLA-4, mCPA (water)

Figure 7: Anti-CTLA-4 delivered by DPX binds to $CD3^+$ T cells even in presence of detected anti-drug antibodies (ADA). Mice treated with DPX-FP/anti-CTLA-4 had similar numbers of circulating $CD3^+$ and $CD8^+$ T cells bound by anti-CTLA-4 (IgG2b) compared to mice treated with systemic anti-CTLA-4 by IP injection (A). Serum samples were collected at 28- and 42-days post-injection from mice in all groups and were assessed for ADA formation by bridging ELISA (B).

Further Information

For further information, contact Marianne Stanford, PhD at mstanford@imv-inc.com

Stability

No degradation in DPX-formulation

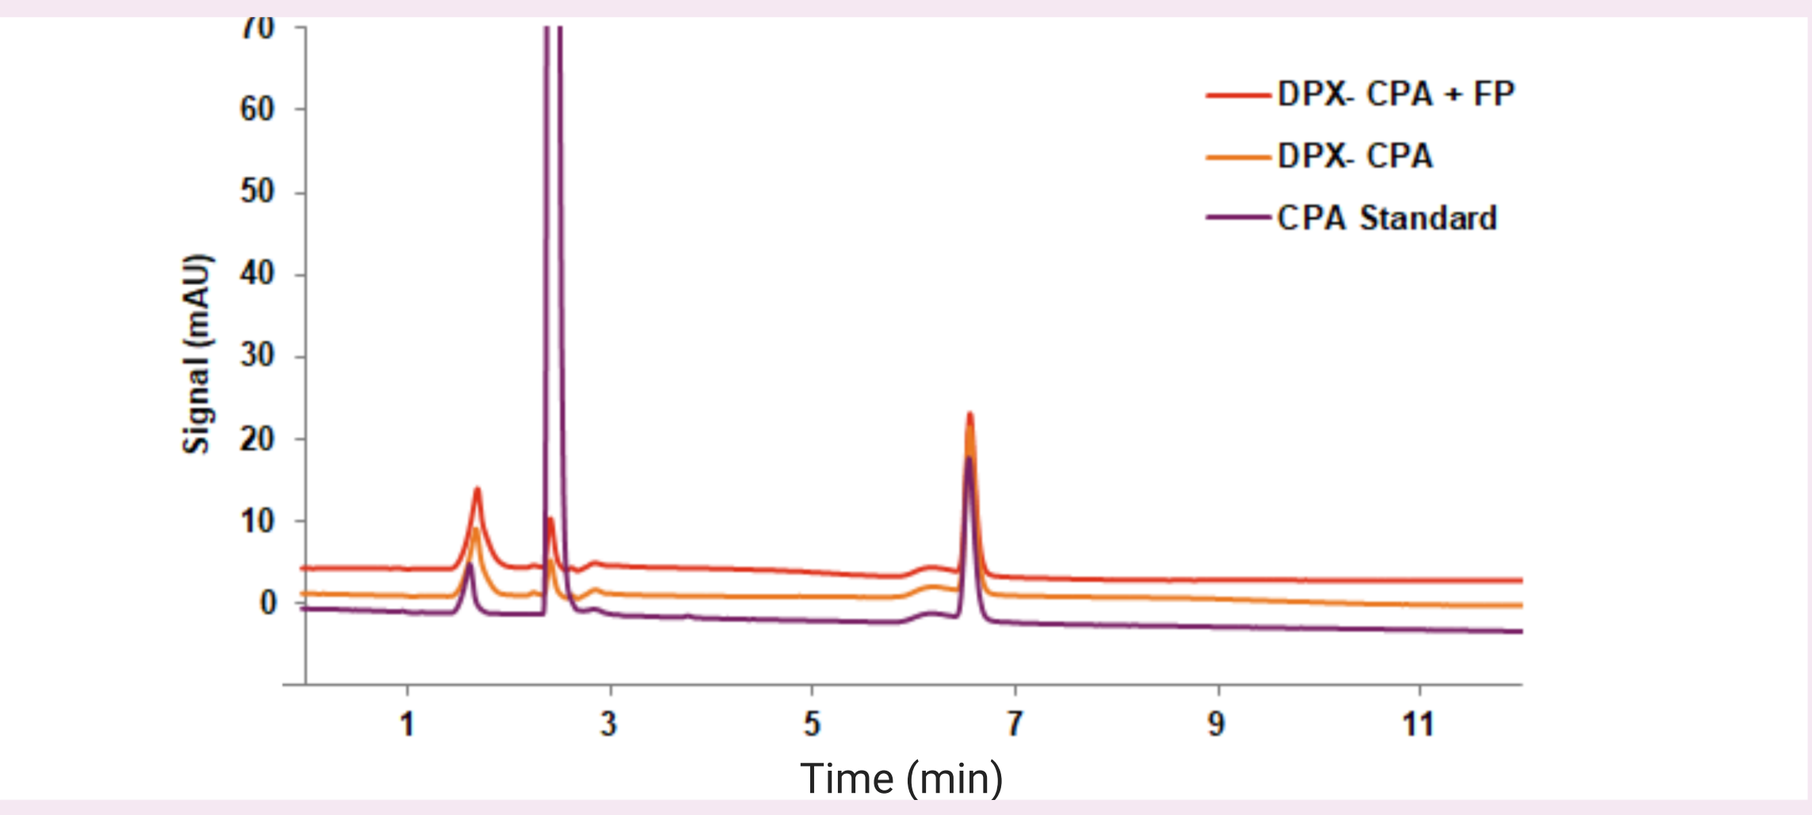
DPX- CPA + FP
DPX- CPA
CPA Standard
Signal (mAU)
Time (min)

Figure 8: Analytical testing of DPX-CPA demonstrates stability of the small molecule drug in DPX-formulations. Chromatogram of high-pressure liquid chromatography (HPLC) displays CPA alone, as DPX-CPA, and as DPX-CPA plus FP. Low doses of CPA were formulated in DPX at 50 µl per dose with 0.04 - 0.4 mg.

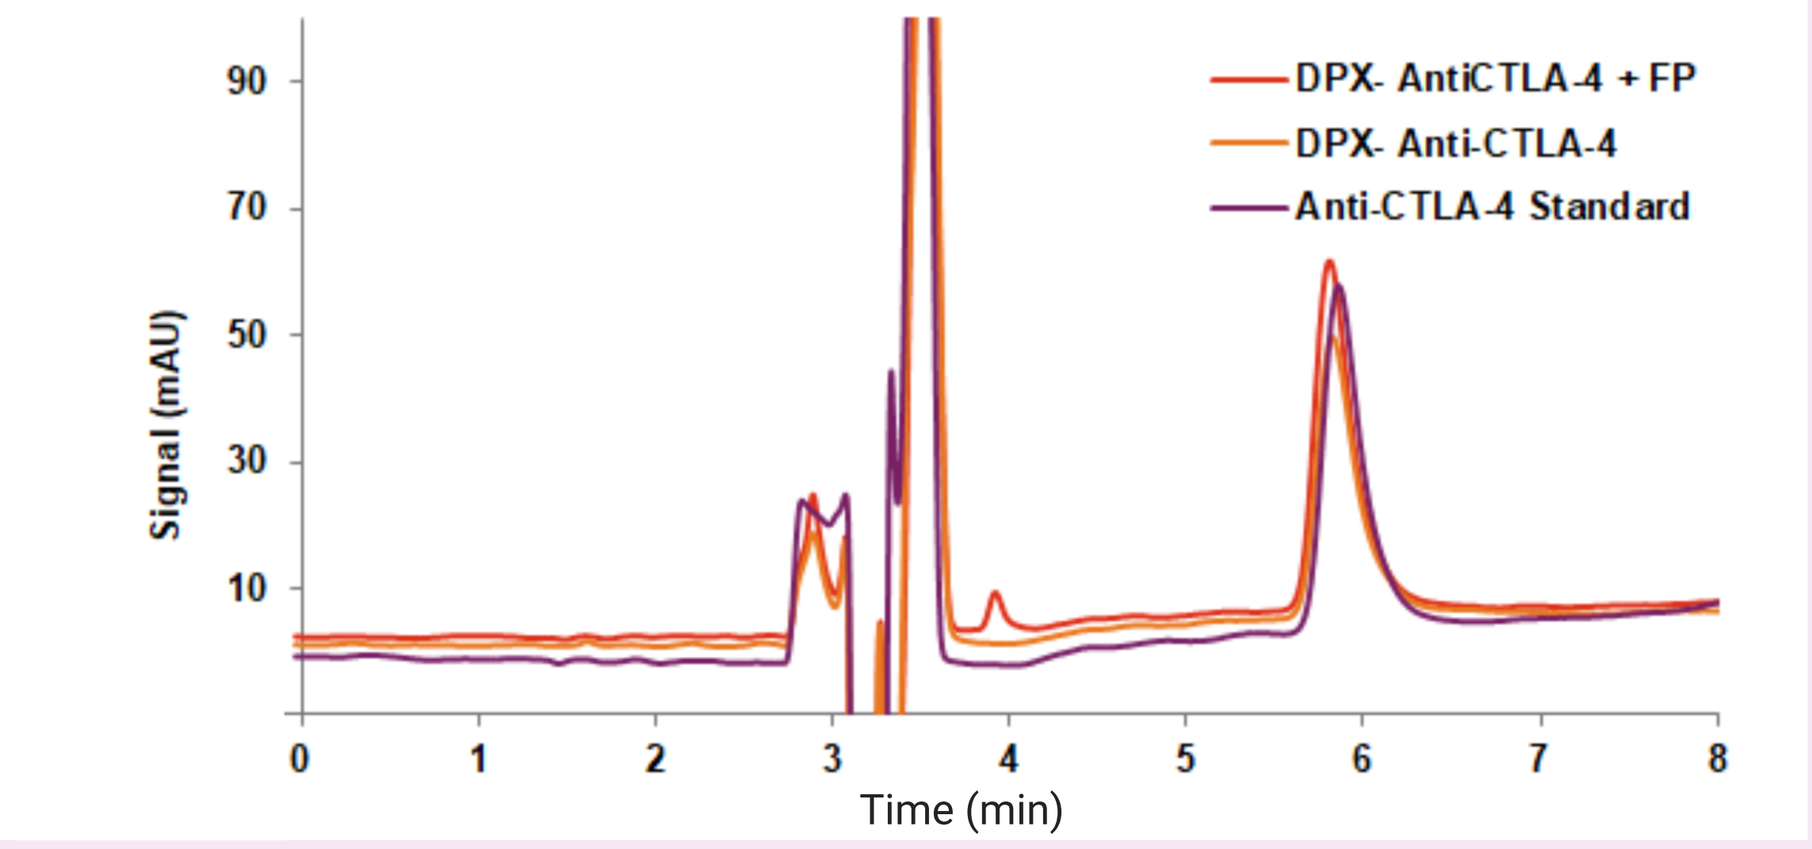
DPX- AntiCTLA-4 + FP
DPX- Anti-CTLA-4
Anti-CTLA-4 Standard
Signal (mAU)
Time (min)

Figure 9: Analytical testing of DPX anti-CTLA-4 demonstrates stability of biological macromolecule in DPX-formulations. HPLC chromatogram shows anti-CTLA-4 alone and as DPX-anti-CTLA-4 with and without FP . Low doses of the checkpoint inhibitor antibody anti-CTLA-4 were formulated in DPX at 50 µl per dose.

Mechanism of Action

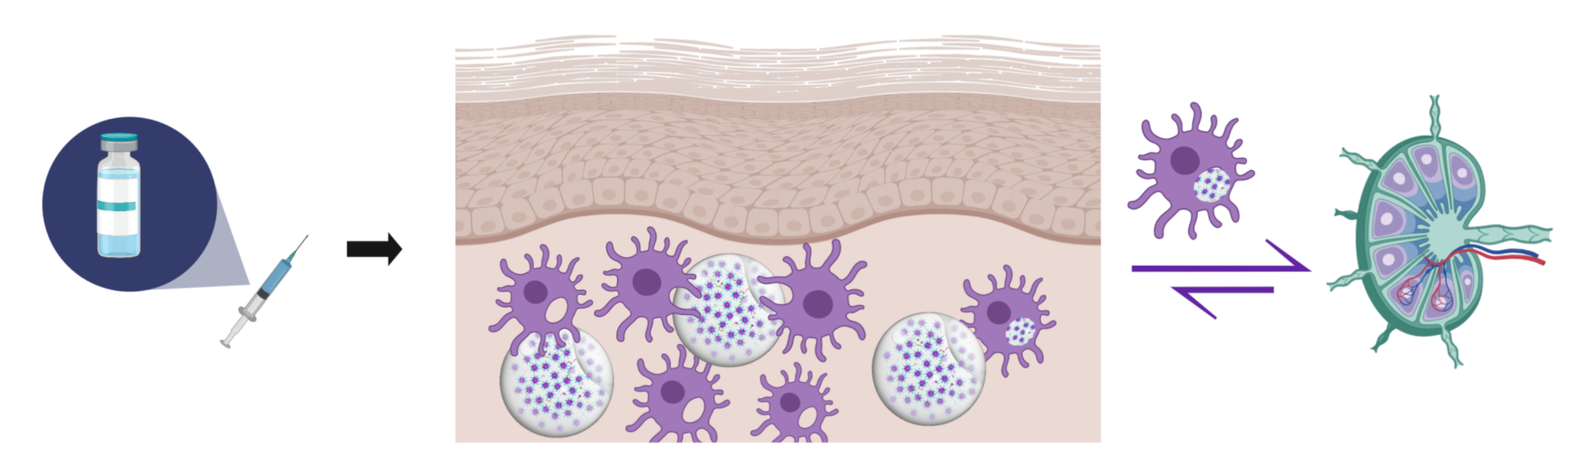

Figure 10: Proposed mechanism of action. DPX-formulations containing immunomodulatory agents, small molecules, and biological macromolecules are injected subcutaneously and carried by dendritic cells to the regional lymph node where they interact directly with immune cells.

CONCLUSIONS

- Targeted delivery of small molecules and immunomodulatory agents to lymph nodes is demonstrated using DPX platform technology.
- DPX-dye experiments demonstrated that DPX can deliver small molecules to lymph nodes with minimal to no systemic exposure.
- Low dose CPA has been shown to act as a lymphodepleting immunomodulatory agent when delivered systemically over a week. As expected, using DPX to deliver CPA resulted in a significant reduction in the number of lymph node cells with significantly less active agent.
- Mice treated with DPX-FP/anti-CTLA-4 developed ADA against anti-CTLA-4. While ADAs may hinder therapeutic efficacy, their generation supports that large biological macromolecular structure is maintained in DPX. Importantly, in this study, the formation of ADAs did not impede the effect of anti-CTLA-4 delivered in DPX in restricting tumour growth nor binding to circulating T cells.