#4098

Monotherapy Activity with the First CD20-Targeted Immunotoxin, MT-3724, in Subjects with Relapsed/Refractory (R/R) Diffuse Large B-Cell Lymphoma (DLBCL)

Paul A. Hamlin, Jr., MD[1], Vasile Musteata, MD, PhD, MPH[2], Mamia Zodelava, MD, PhD[3], Steven I. Park, MD[4], Christine Burnett, PhD[5], Kristina Dabovic, PharmD[5], Eric T. Williams, PhD[5], Jack P. Higgins, PhD[5], Daniel O. Persky, MD[6]

[1]Department of Medicine, Lymphoma Service, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Institute of Oncology, ARENSIA Exploratory Medicine Unit, Chisinau, Republic of Moldova; [3]ARENSIA Exploratory Medicine LLC, Tblisi, Georgia; [4]Levine Cancer Institute, Charlotte, NC, USA; [5]Molecular Templates, Austin, TX, USA; [6]University of Arizona Cancer Center, Tucson, Arizona, USA

Background

- The near ubiquity and persistence of CD20 expression in B-cell malignancies provides a strong rationale for novel CD20-directed therapies[1]
- MT-3724 represents a novel engineered toxin body (ETB) modality comprised of an anti-CD20 single-chain variable fragment genetically fused to Shiga-like Toxin A subunit (SLTA); MT-3724 is capable of efficient internalization once bound to CD20 and can induce potent direct cell-kill via enzymatic ribosome inactivation (**Figure 1**)[2]

Figure 1. Internalization of MT-3724 and Ribosome Inactivation of Target Cell

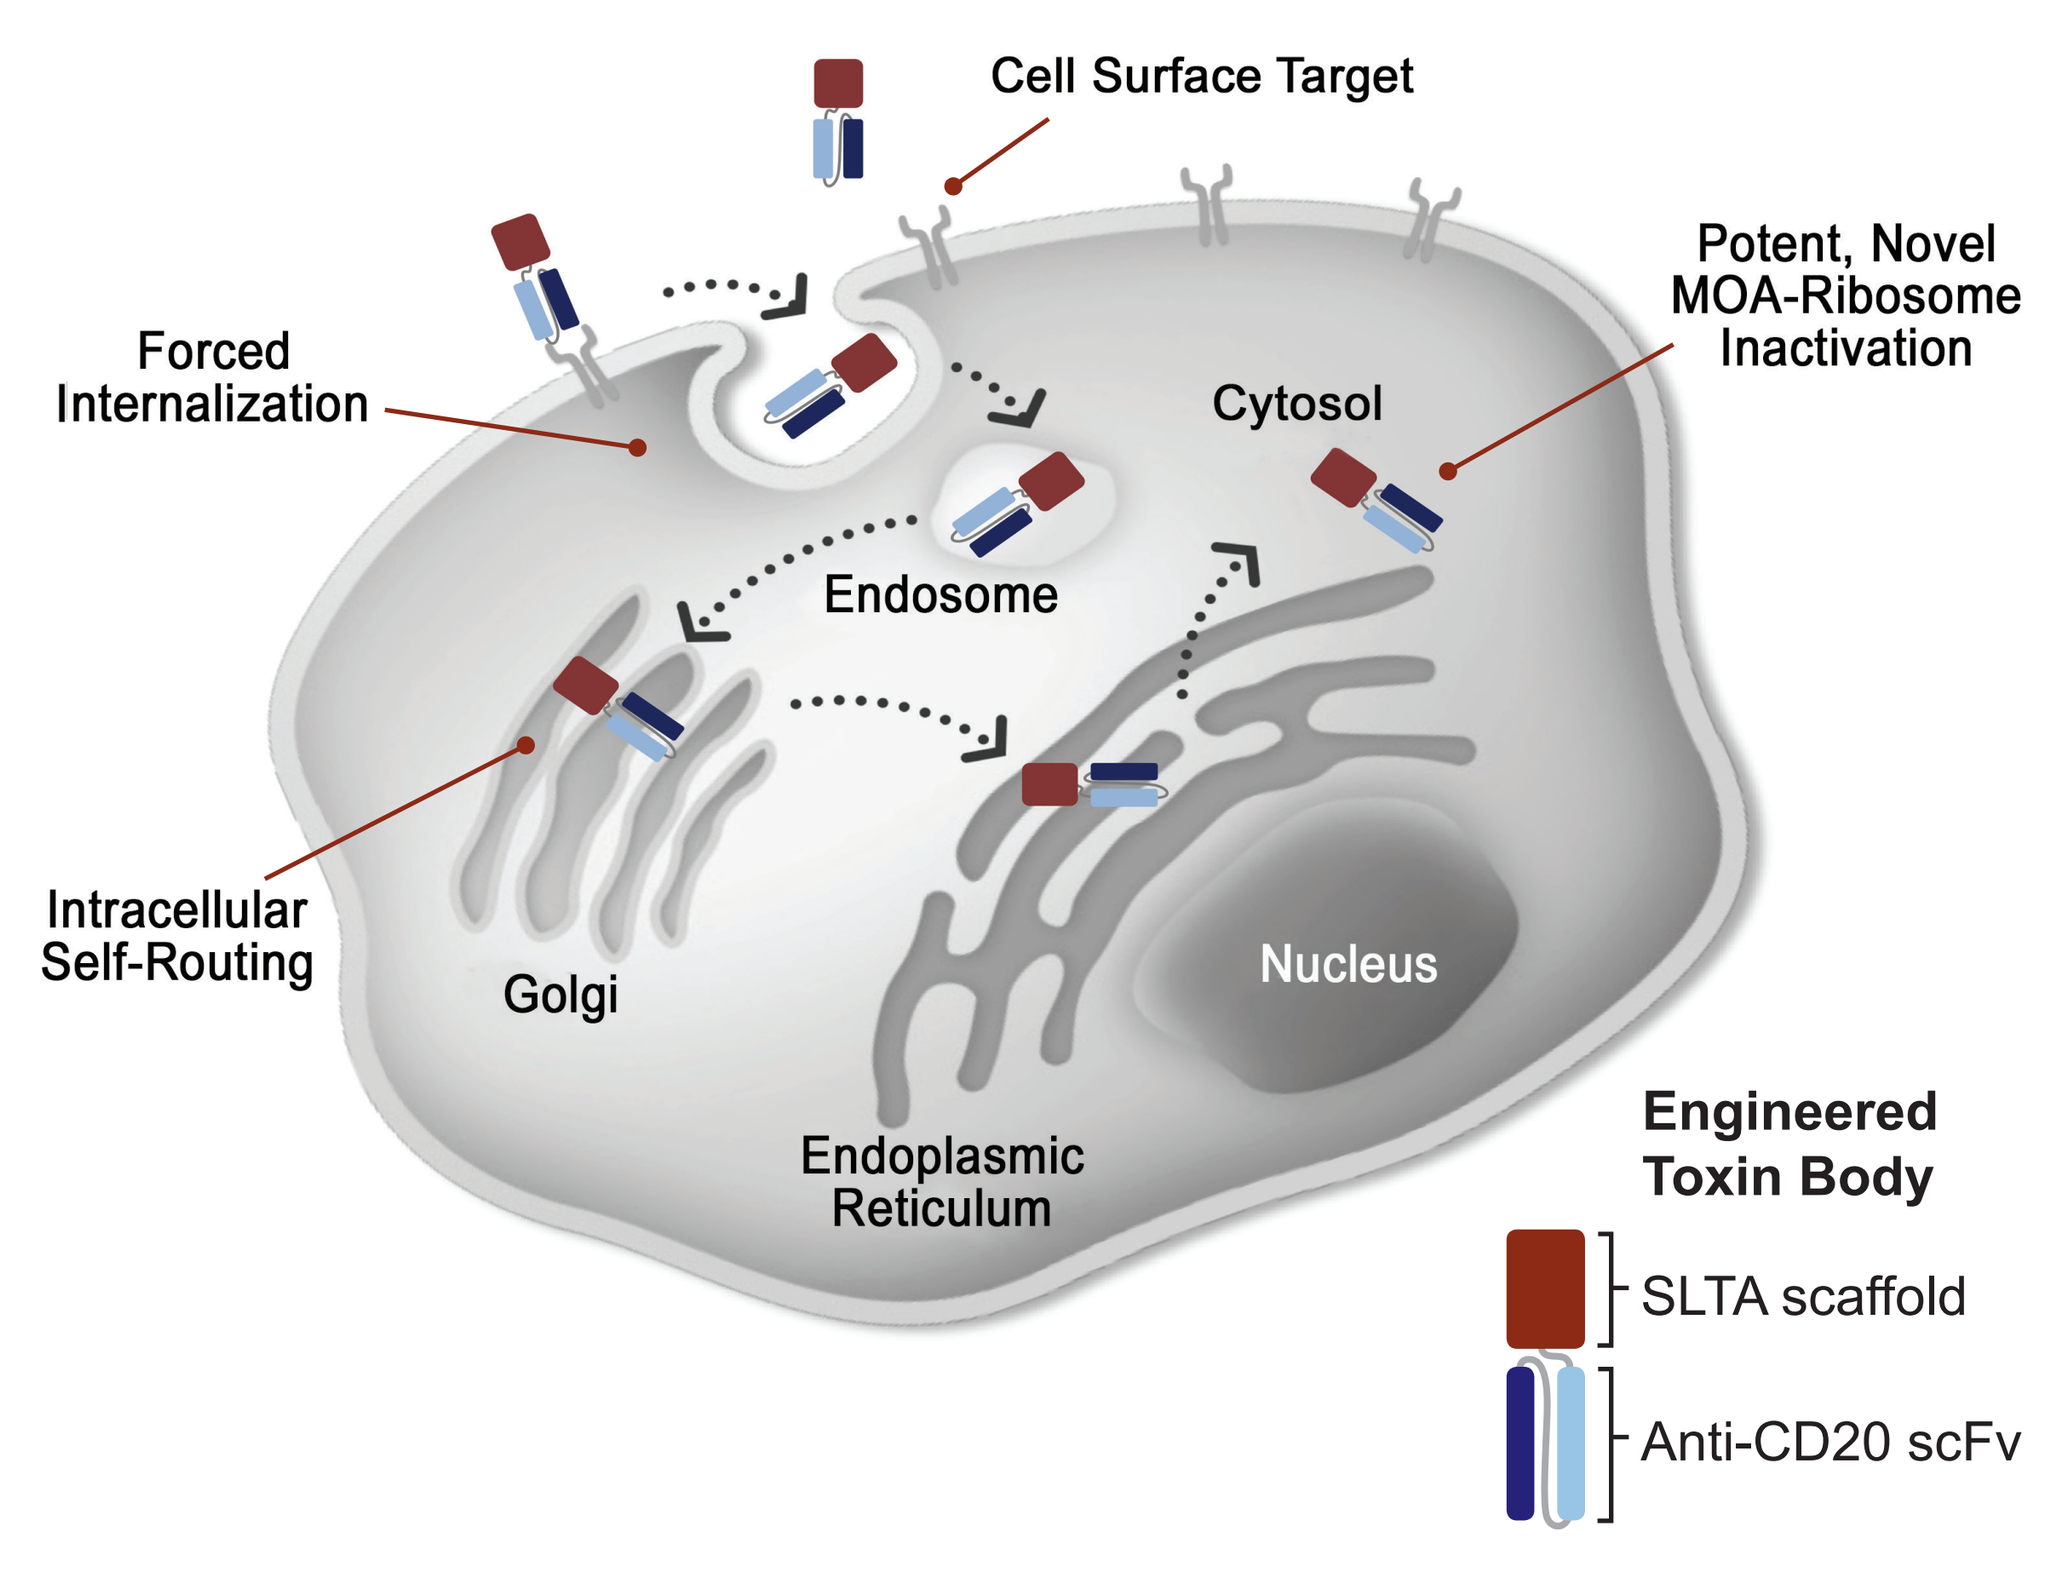
Cell Surface Target
Forced Internalization
Cytosol
Potent, Novel MOA-Ribosome Inactivation
Endosome
Intracellular Self-Routing
Golgi
Nucleus
Endoplasmic Reticulum
Engineered Toxin Body
SLTA scaffold
Anti-CD20 scFv

MOA = mechanism of action; scFv = single-chain variable fragment; SLTA = Shiga-like toxin A.

Objective

- Present safety and efficacy results from dose escalation to maximum tolerated dose (MTD) in non-Hodgkin lymphoma (NHL) subjects and dose expansion in DLBCL subjects

Methods

- This is a multi-center, open-label, multiple-dose, dose-escalation and dose-expansion study of MT-3724 (NCT02361346)
- The phase 1/1b portion of the study consists of Parts 1 and 2; in addition, a phase 2 dose-expansion portion of the study (Part 3) has been initiated and is open to recruitment (**Figure 2**):
 - Part 1: The MT-3724 dose was escalated in 21 subjects with B-cell NHL according to a 3+3 design based on ≤1 subject exhibiting dose-limiting toxicity (DLT) during Cycle 1 (28 days for first cycle, 21 days for subsequent cycles) in 6 sequential-dose cohorts (5, 10, 20, 50, 100 and 75 µg/kg/dose IV 3 times per week for the first two weeks of each cycle) for up to 5 cycles
 - Part 2: Safety and efficacy of MT-3724 were further evaluated in 6 serum rituximab-negative (RTX-neg) subjects with DLBCL; tumor response was assessed by the International Working Group Response Criteria for Clinical Trials[3]
 - For Parts 1 and 2: Retreatment under a separate extension protocol was considered for subjects who exhibited partial response (PR) or stable disease (SD) after completing 5 cycles (or 2 cycles after a complete response [CR])
 - Part 3: This is a multi-center, multi-national, phase 2 open-label, single-arm evaluation of MT-3724 given as monotherapy in repeat doses in subjects with R/R DLBCL who have received 2 or more lines of prior therapy; this part includes three stages, where the first two stages will follow the Simon two-stage optimum design[4]

Figure 2. Study Schema

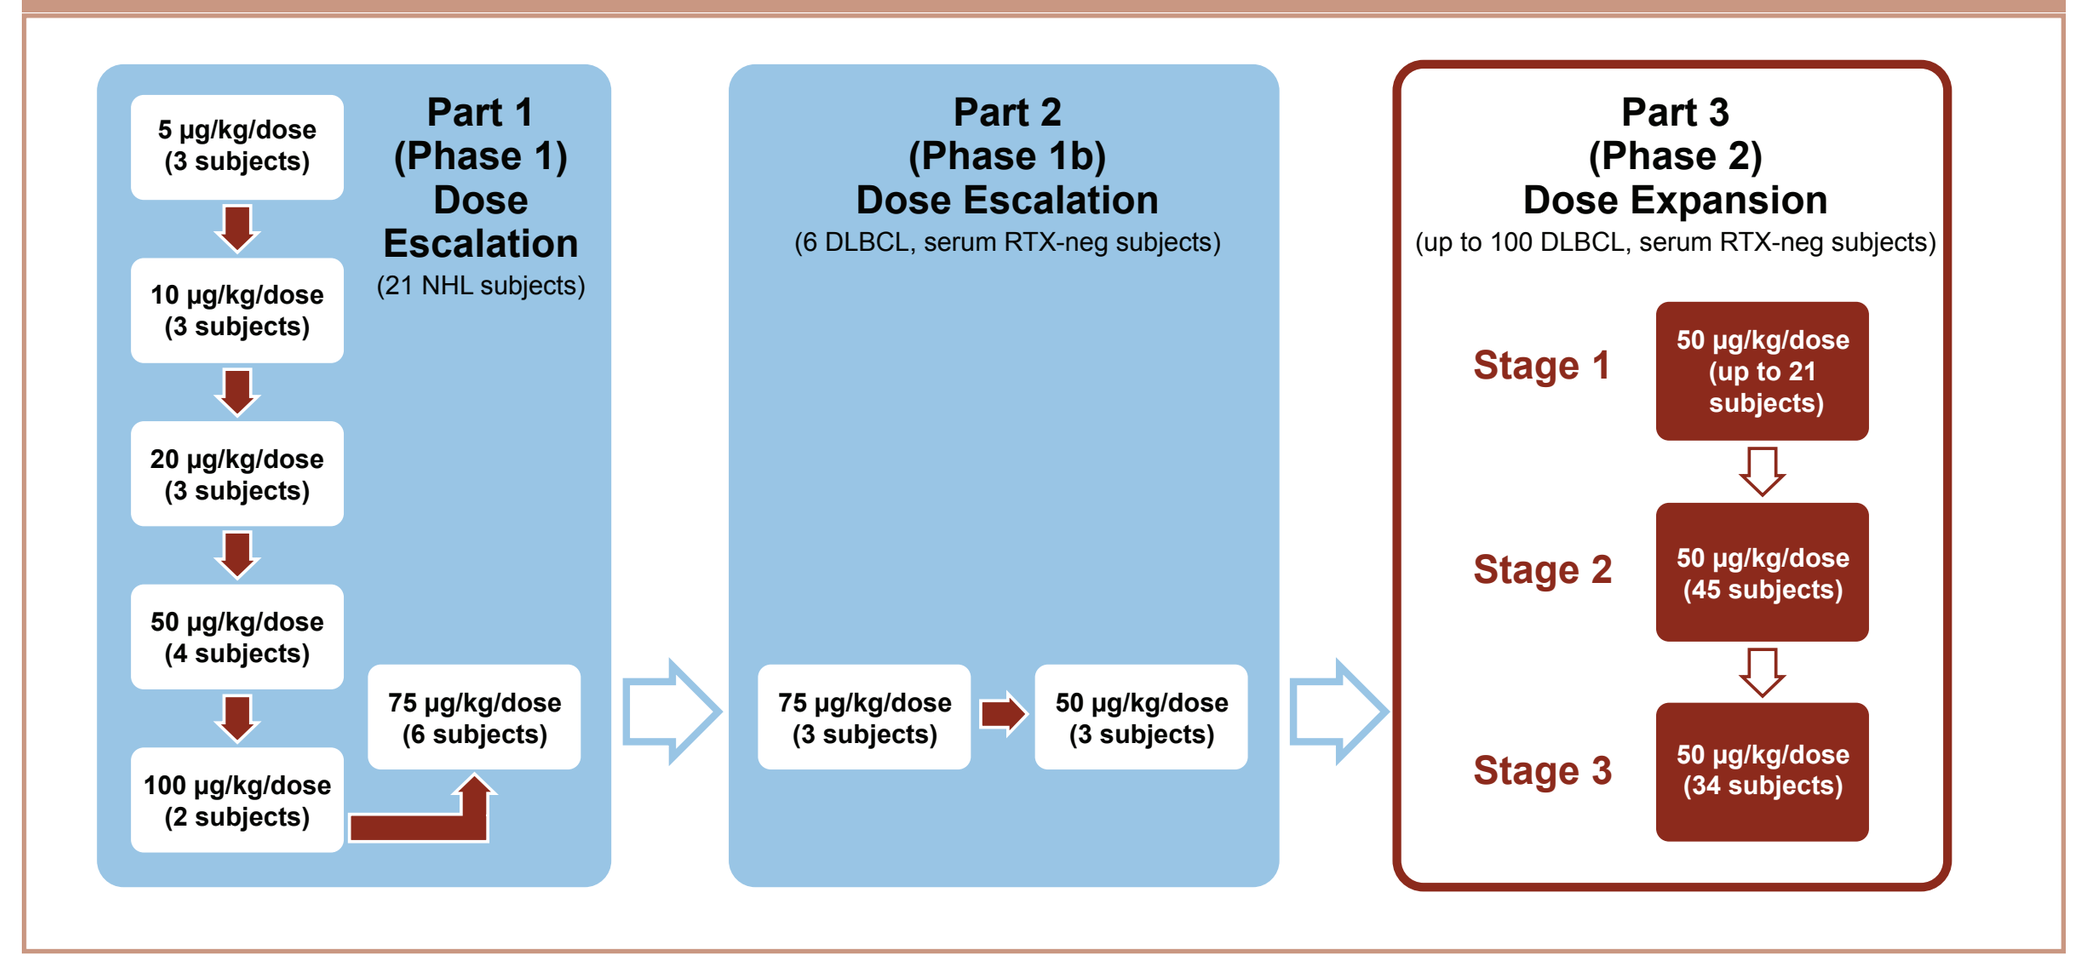
5 µg/kg/dose (3 subjects)
10 µg/kg/dose (3 subjects)
20 µg/kg/dose (3 subjects)
50 µg/kg/dose (4 subjects)
100 µg/kg/dose (2 subjects)
75 µg/kg/dose (6 subjects)
Part 1 (Phase 1) Dose Escalation
(21 NHL subjects)
Part 2 (Phase 1b) Dose Escalation
(6 DLBCL, serum RTX-neg subjects)
75 µg/kg/dose (3 subjects)
50 µg/kg/dose (3 subjects)
Part 3 (Phase 2) Dose Expansion
(up to 100 DLBCL, serum RTX-neg subjects)
Stage 1
50 µg/kg/dose (up to 21 subjects)
Stage 2
50 µg/kg/dose (45 subjects)
Stage 3
50 µg/kg/dose (34 subjects)

- Key eligibility criteria include:
 - B-cell NHL/chronic lymphocytic leukemia (CLL)
 - NHL: intermediate or high risk, as determined by International Prognostic Index (IPI)/Follicular Lymphoma International Prognostic Index (FLIPI); CLL: Rai Stage III/IV
 - DLBCL only in Parts 2 and 3
 - Relapse after measurable response to at least one anti-CD20 containing regimen
 - Exhausted all approved therapies
 - Eastern Cooperative Oncology Group (ECOG) Performance Status (PS) 0 to 2
 - Typical hematologic and chemistry parameters for a phase 1/2 trial
 - A minimum of 12-week washout is required after last course of RTX
 - In Part 1, all subjects' serum RTX levels were assessed but this was not part of eligibility
 - Following a mixed response in only 1 of 7 subjects with measurable serum RTX levels who were evaluable for response, Part 2 eligibility was amended to require a serum RTX-neg assessment at study entry

Results

- Twenty-seven subjects with NHL were enrolled; baseline characteristics are shown in **Table 1**
 - 21 in Part 1 (dose escalation) – completed
 - 6 in Part 2 (expansion cohort) – closed to enrollment
- Median duration of treatment was 39 (5-362) days

Table 1. Baseline Characteristics

Characteristic	N=27
Sex	
F	17
M	10
Age (mean, range; years)	65 (34 – 78)
Body weight (mean, range; kg)	81 (51 – 154)
BMI (median, range; kg/m²)	29.1 (19 – 44.6)
ECOG performance status	
0	10
1	13
2	4
NHL type[a]	
DLBCL	16
Mixed DLBCL/FL	4[b]
FL	5
MCL	2
Prior NHL treatments (median, range)	4 (1 – 11)
Prior anti-CD20 treatments (median, range)	2 (1 – 9)
RTX level in serum	
<500 ng/mL	19[c]
≥500 ng/mL	8

DLBCL = diffuse large B-cell lymphoma; FL = follicular lymphoma; MCL = mantle cell lymphoma; NHL = non-Hodgkin lymphoma; RTX = rituximab.
[a]Although eligible for this study, no subject with CLL was enrolled. [b]One subject transformed from DLBCL to FL following auto-stem cell transplant (SCT) prior to study entry. [c]One subject did not require testing; >365 days since last RTX.

Maximum Tolerated Dose

- Expansion cohort was open for enrollment at 5 sites: US (2), Canada (1) and Eastern Europe (2)
 - MT-3724 was not tolerated at 100 µg/kg/dose (DLTs: grade 3 pneumonia, grade 2 ileus)
- The MTD was initially defined at 75 µg/kg/dose
- The first 3 subjects treated in Part 2 at 75 µg/kg in the expansion cohort exhibited medically important adverse events (AEs) related to MT-3724
 - 2 subjects (both with BMI >35) had grade 2 capillary leak syndrome (CLS); both subjects started at 75 µg/kg (total dose of 11,573 µg/infusion and 7,207 µg/infusion, respectively), with this dose reduced to 50 µg/kg after C1D2 and C1D1, respectively
 - The third subject had a serious AE (SAE) of grade 3 edema
- The MTD was reduced to 50 µg/kg/dose, with an upper limit of 6000 µg/dose
- 3 subjects have now been treated in Part 2 at 50 µg/kg/dose with the upper limit of 6000 µg/dose
 - 1 subject received 9 full cycles and achieved a CR; data are still being collected

Safety

- The most common treatment-emergent AEs (TEAEs) of any grade were peripheral edema, fatigue, diarrhea and myalgia (**Table 2**)

Table 2. Treatment-Emergent Adverse Events Occurring in >15% Subjects

TEAE	N=27 n (%)	TEAE	N=27 n (%)
Peripheral edema	17 (63)	Hypotension	6 (22)
Fatigue	11 (41)	Anemia	5 (19)
Diarrhea	11 (41)	Stomatitis	5 (19)
Myalgia	11 (41)	Asthenia	5 (19)
Insomnia	8 (30)	Pneumonia	5 (19)
Nausea	7 (26)	Hypoalbuminemia	5 (19)
Pyrexia	7 (26)	Hypokalemia	5 (19)
Cough	7 (26)	Dyspnea	5 (19)
Arthralgia	6 (22)	Rash	5 (19)
Headache	6 (22)		

- 27 subjects had TEAEs with related causality, the most common were: peripheral edema (44%), myalgia (30%), fatigue (26%), nausea (26%), diarrhea (22%), pyrexia (22%), headache (19%) and stomatitis (19%)
- The most common related grade ≥3 TEAEs included neutropenia (including neutrophil count decrease) and myalgia (each 3/27 [11%])
- There were 9 related SAEs among 6 subjects (**Table 3**)

Table 3. Serious Adverse Events Attributable to MT-3724

Subject	SAE	Grade	Length of Hospitalization	Outcome	Subject Disposition
100 µg/kg					
14	Ileus[a]	2	9 days	Resolved	Withdrawn AE
	Muscle weakness	3	9 days	Resolved	
15	Pneumonia[a]	3	1 day	Resolved	Withdrawn AE
75 µg/kg					
19	Peripheral Edema	2	7 days	Ongoing	Withdrawn PD
23	Edema	3	14 days	Resolved	Withdrawn PD
	Pneumonia	3	14 days	Resolved	
20	Acute Kidney Injury (AKI)	4	12 days	Ongoing	Withdrawn AE (AKI)
	Viral Syndrome	2	3 days	Resolved	
50 µg/kg					
13	Worsening Hypertension	3	1 day	Resolved	Withdrawn AE

[a]Dose-limiting toxicity.

- Two of the subjects treated had prior CAR-T therapy and had no Grade >1 AEs considered related to MT-3724
- One subject died on study from disease progression
- Overall, AEs expected to be related to MT-3724 due to reducing normal lymphocytes (eg, infections), or eliciting innate immune responses (eg, CLS, myalgia, arthralgia), were observed; however, no cases of Grade >2 CLS were observed

Efficacy

- The maximal reduction of B-cells (nadir) from 5 subjects who were serum RTX-neg at study initiation, who completed 5 cycles of MT-3724 treatment, and with >10 (cells/µL) CD19+ peripheral B-cells at baseline are shown in **Figure 3**

Figure 3. Dose-Dependent Peripheral B-Cell Depletion by MT-3724 Dose (Maximal Change From Baseline)

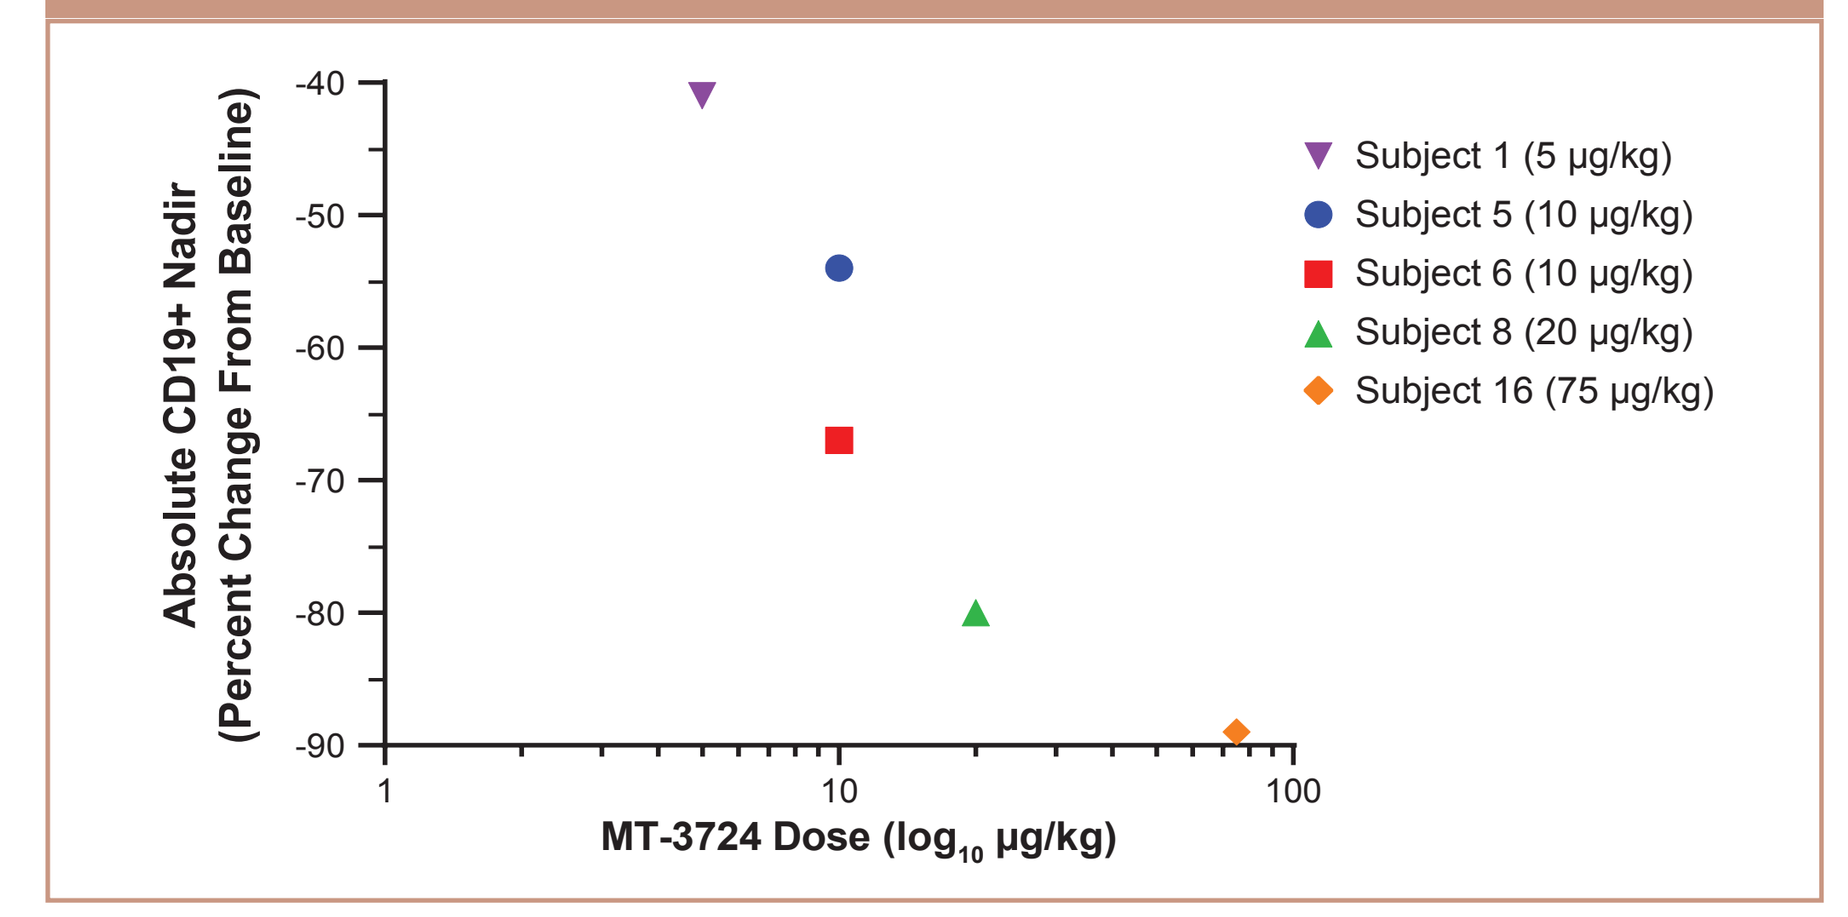
Absolute CD19+ Nadir (Percent Change From Baseline)
-40
-50
-60
-70
-80
-90
1
10
100
MT-3724 Dose (log_{10} µg/kg)
Subject 1 (5 µg/kg)
Subject 5 (10 µg/kg)
Subject 6 (10 µg/kg)
Subject 8 (20 µg/kg)
Subject 16 (75 µg/kg)

- In total, 25 subjects were evaluable for efficacy, including 19 subjects with DLBCL or mixed DLBCL/follicular lymphoma (FL); 2 subjects were not evaluable for efficacy (**Figure 4**)
- Of the 13 serum RTX-neg DLBCL or mixed DLBCL/FL subjects, 5 (38% objective response rate) responded across the range of 5 to 50 µg/kg/dose (**Figure 5**)**; of subjects with RTX-neg DLBCL who received the MTD of 50 µg/kg/dose, 3 of 5 responded**
 - Of the 5 responses, 2 were CRs and 3 were PRs, including Subject 8 with composite DLBCL/FL post-autologous stem cell transplantation (post-SCT) who had a complete metabolic response of a large mesenteric mass and proceeded to allogeneic SCT
 - Among the 5 responders, 2 subjects with CR discontinued (subject decision) due to reasons unrelated to safety events, and durations of response were 169 and 90 days; 3 subjects with PR discontinued after disease progression, and durations of response were 107, 120, and 75 days
 - Of the responders, median age was 61 (50-76) yrs, and median number of prior lines of NHL therapy received was 3 (1-8)
 - Three serum RTX-neg subjects with DLBCL or mixed DLBCL/FL had SD, including two with 49% and 47% reductions in index tumor lesions (**Figure 5**)
- Of all 25 subjects evaluated for efficacy, 13 developed anti-drug antibodies (ADAs) or neutralizing antibodies (NAbs); ADAs/NAbs were observed in 4 of 5 subjects who showed a response, and in 3 of 5 subjects with SD; some subjects even had improvements in response following development of ADAs and NAbs (**Figure 4**)
- Three of the 4 subjects with detectable baseline serum RTX levels who were evaluable for response did not benefit from MT-3724; the fourth subject experienced a decrease in index lesions but an increase in non-target lesions as well as new lesions (**Figure 5**)

Figure 4. Objective Response, Development of ADAs/NAbs and Time on Study

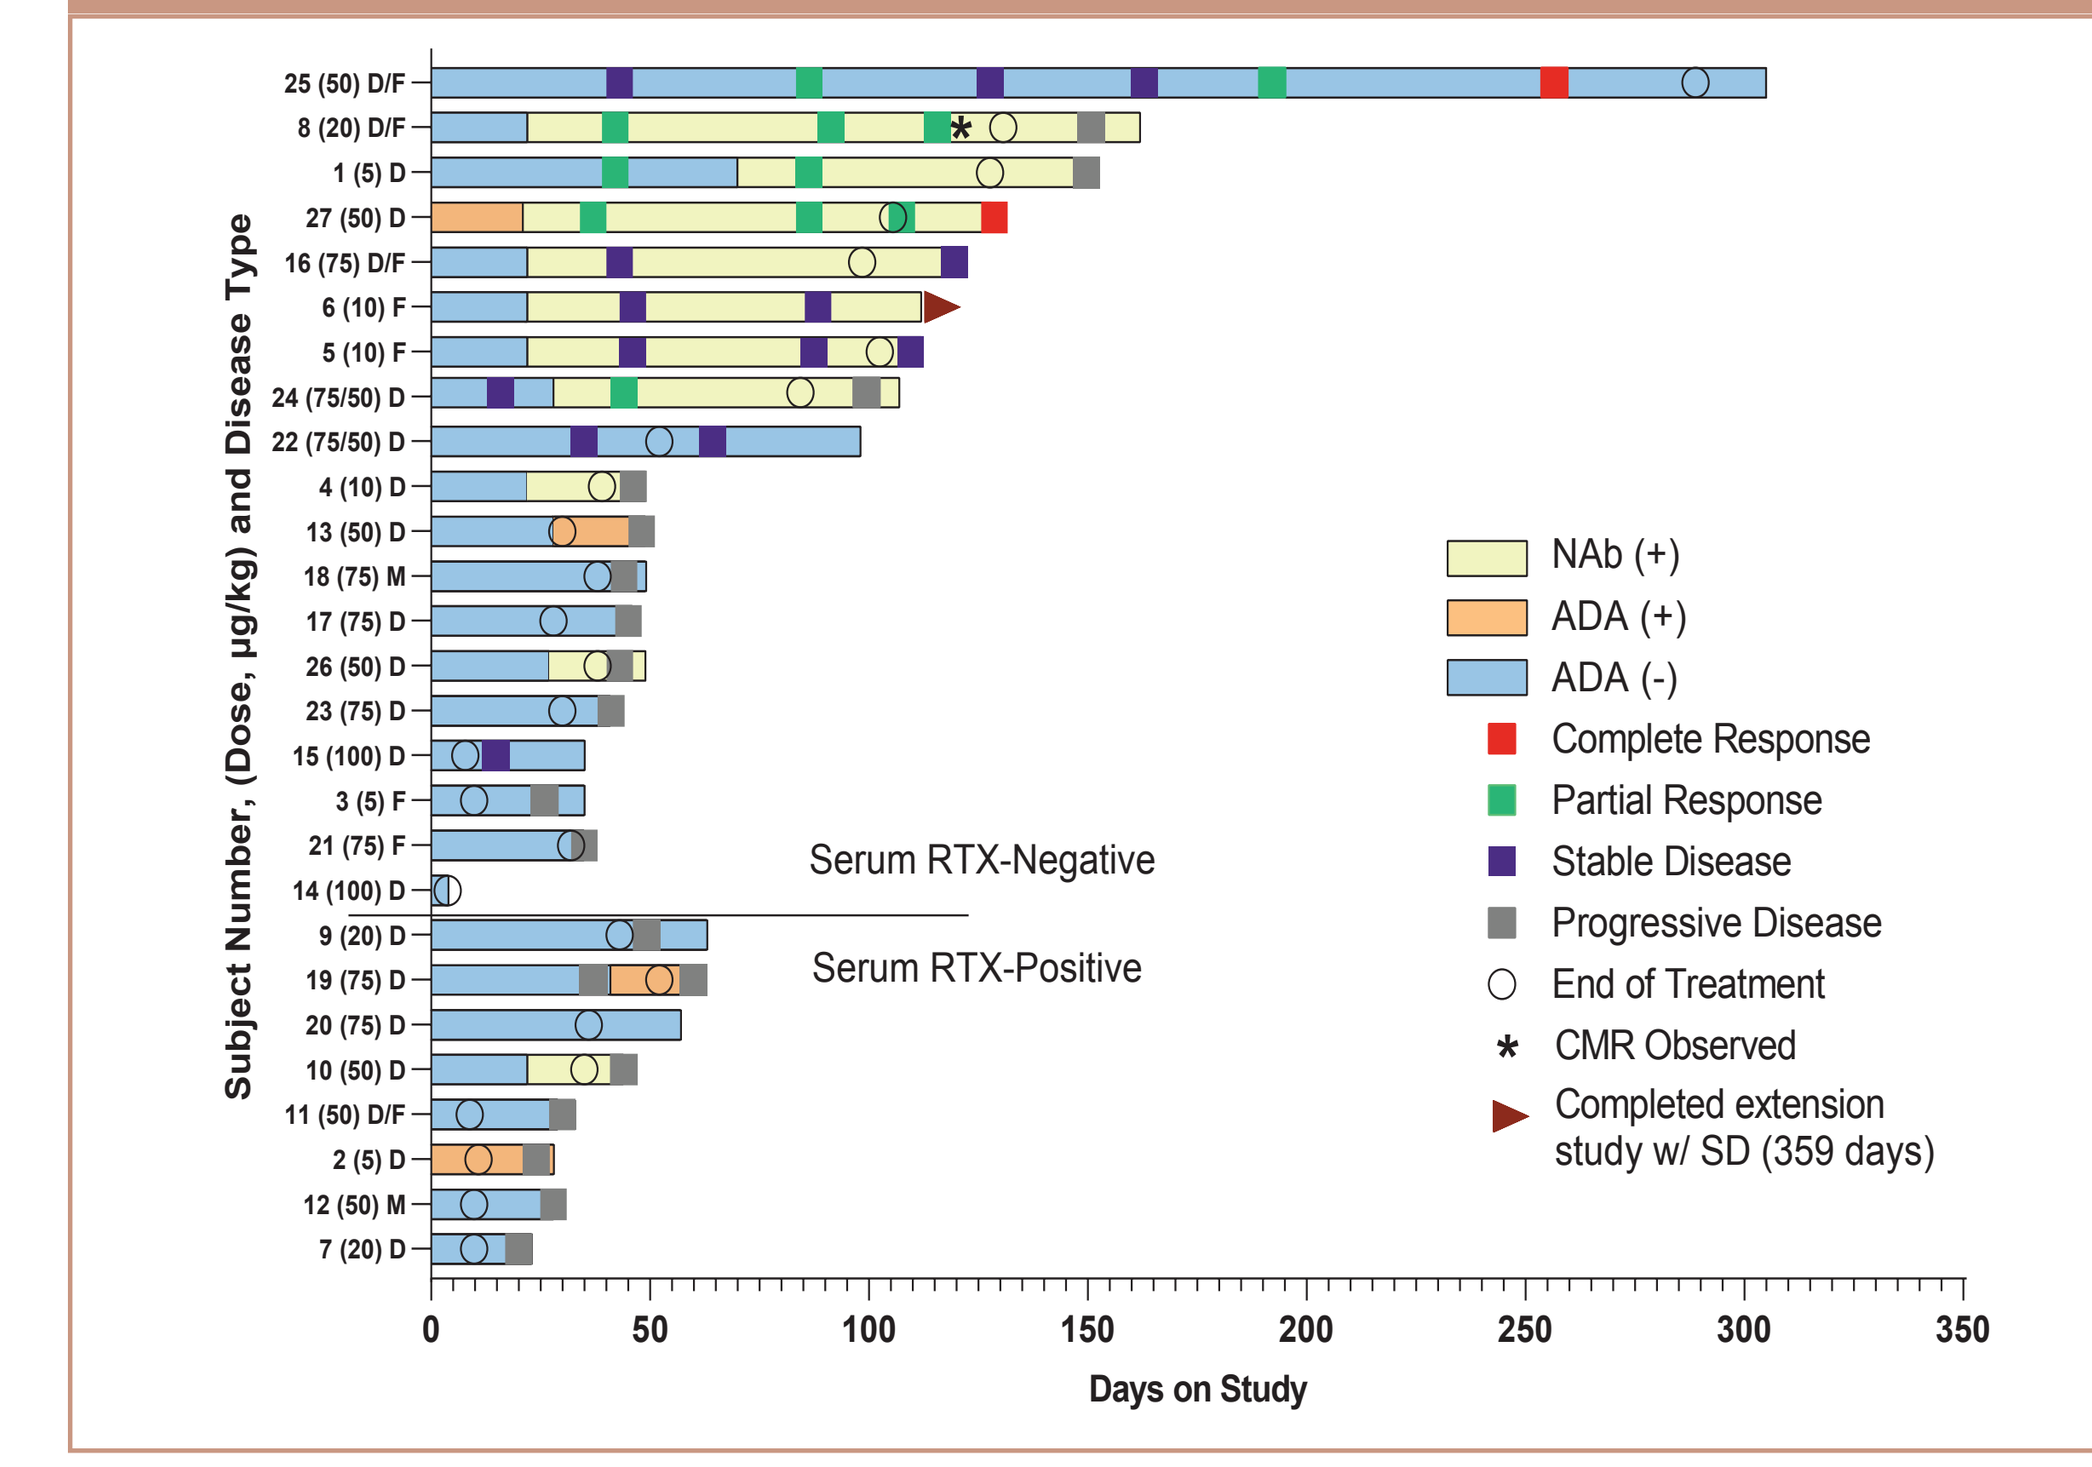
Subject Number, (Dose, µg/kg) and Disease Type
25 (50) D/F
8 (20) D/F
1 (5) D
27 (50) D
16 (75) D/F
6 (10) F
5 (10) F
24 (75/50) D
22 (75/50) D
4 (10) D
13 (50) D
18 (75) M
17 (75) D
26 (50) D
23 (75) D
15 (100) D
3 (5) F
21 (75) F
14 (100) D
9 (20) D
19 (75) D
20 (75) D
10 (50) D
11 (50) D/F
2 (5) D
12 (50) M
7 (20) D
Serum RTX-Negative
Serum RTX-Positive
NAb (+)
ADA (+)
ADA (-)
Complete Response
Partial Response
Stable Disease
Progressive Disease
End of Treatment
CMR Observed
Completed extension study w/ SD (359 days)
0
50
100
150
200
250
300
350
Days on Study

ADA = anti-drug antibody; CMR = complete metabolic response (best response was PR with CMR observed in the largest lesion); CR = complete response; D = diffuse large B-cell lymphoma; F = follicular lymphoma; M = mantle cell lymphoma; NAb = neutralizing antibody; PD = progressive disease; PR = partial response; RTX = rituximab; SD = stable disease.
Subjects 14 and 20 did not have a response assessment; subject 14 left the study due to a dose-limiting toxicity; subject 20 left study due to an adverse event.
Subject 6 entered the extension study after a 3-month break, and the subject left the study with SD.
Subject 8 completed 1 cycle of the extension study and went on to receive allogeneic SCT post-study.
Subject 24 dose reduced from 75 to 50 µg/kg after 1 dose; subject 22 dose reduced from 75 to 50 µg/kg after 2 doses.

Figure 5. Percent Change of Target Lesions

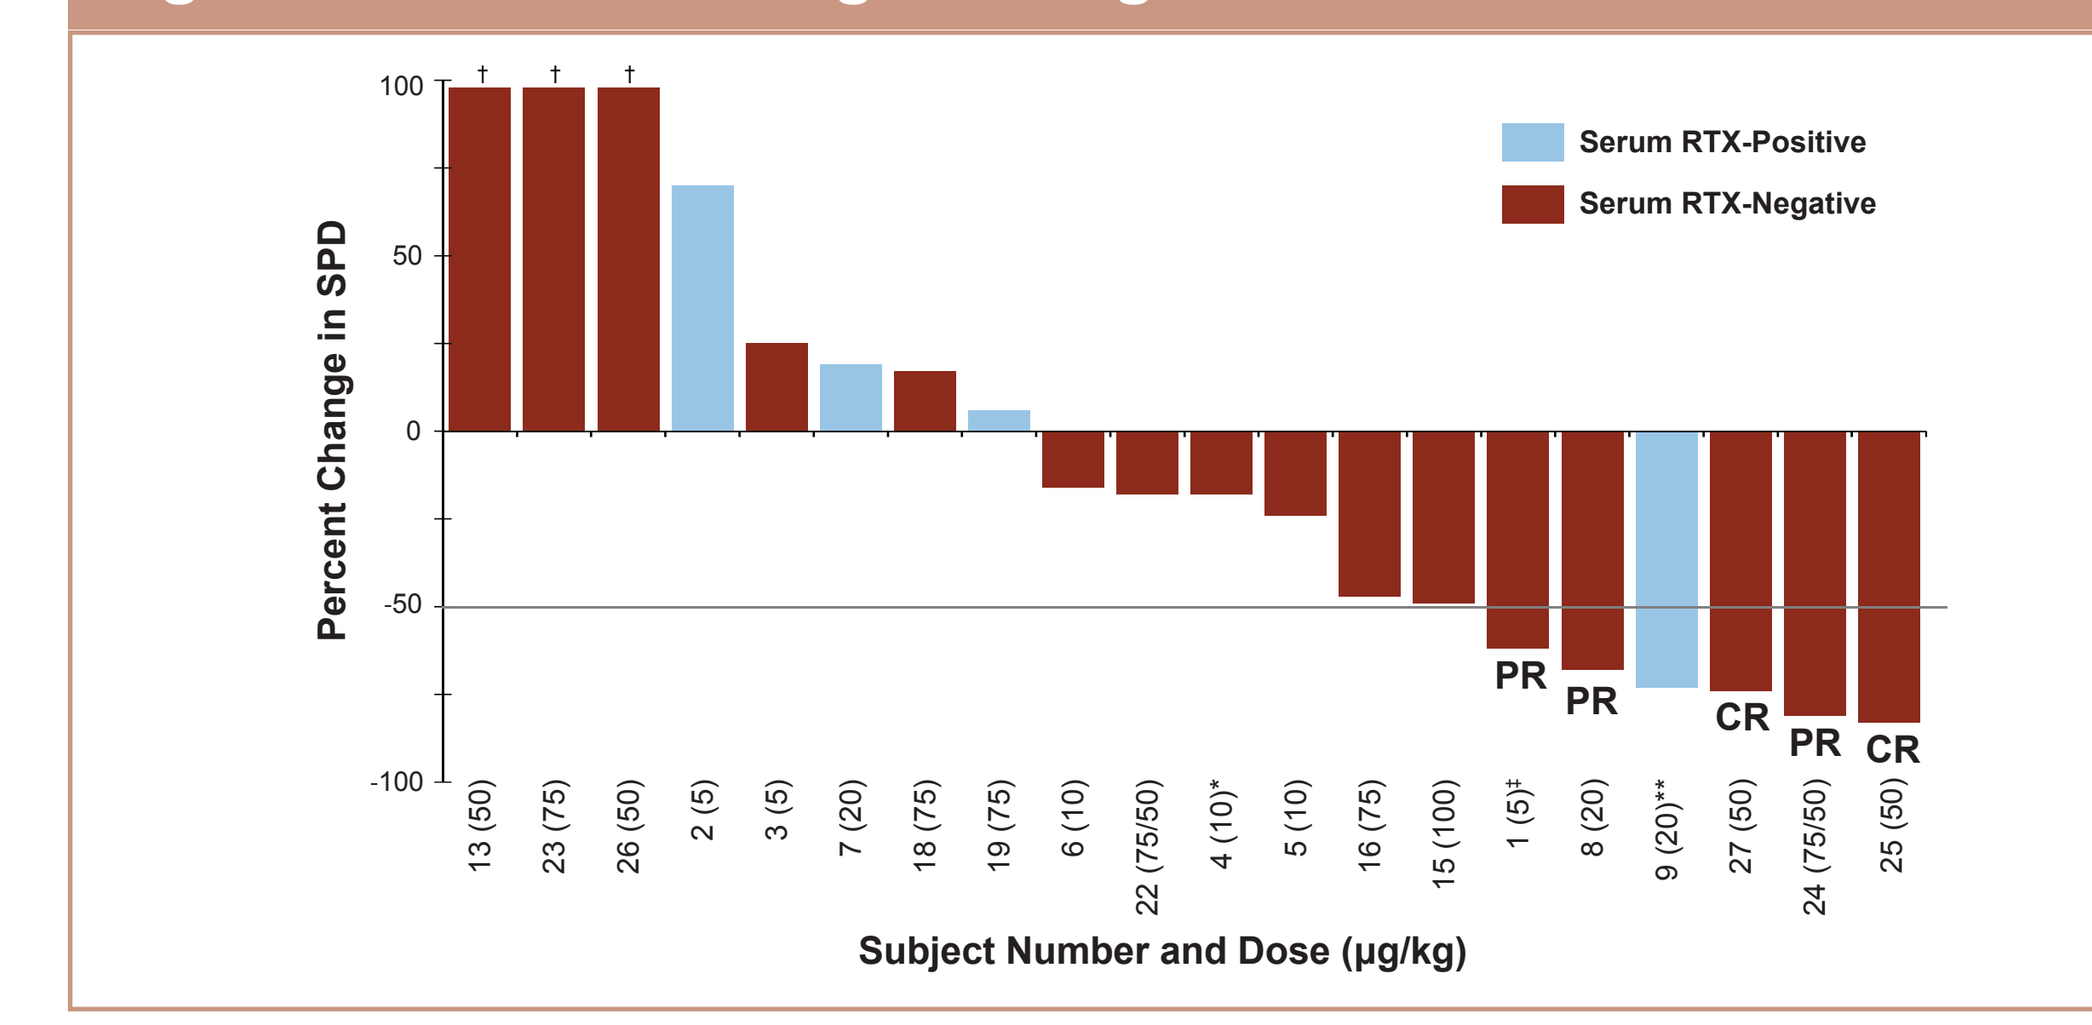
Percent Change in SPD
100
50
0
-50
-100
Serum RTX-Positive
Serum RTX-Negative
†
†
†
PR
PR
CR
PR
CR
13 (50)
23 (75)
26 (50)
2 (5)
3 (5)
7 (20)
18 (75)
19 (75)
6 (10)
22 (75/50)
4 (10)*
5 (10)
16 (75)
15 (100)
1 (5)‡
8 (20)
9 (20)**
27 (50)
24 (75/50)
25 (50)
Subject Number and Dose (µg/kg)

CR = complete response; PR = partial response; RTX = rituximab; SPD = sum of the product of the diameters.
One serum RTX-positive subject (subject number 10) had incomplete evaluation of index lesions following Cycle 1 but an apparent increase of 11% in SPD and subsequently died during Cycle 2 due to progressive disease. In addition, 6 subjects did not have tumor response assessed radiographically (3 RTX-positive [subject numbers 11, 12, 20; 2 discontinued for clinical progression and one discontinued for AE, respectively] and 3 RTX-negative [subject numbers 17, 21, 14; 2 discontinued for clinical progression and one discontinued for AE, respectively]).
†Subjects 13, 23 and 26 had % change in SPD >100%, as follows: 201%, 182% and 304%, respectively.
*PD: Progression of non-target lesions.
**PD: 73% reduction in target lesions post Cycle 2 but substantial increase in non-target lesions and new lesions detected.
‡PR: Best response 62% reduction in target lesions. Progression at end of study due to progression of non-target lesions.

CONCLUSIONS

- MT-3724 is the first CD20-targeted immunotoxin to enter clinical trials
- Safety events were mostly mild to moderate, and DLTs were indicative of innate immune response
- A tolerable dose schedule for phase 2 has been identified: 50 µg/kg/dose up to a maximum of 6000 µg/dose infused over 1 hour on Days 1, 3, 5, 8, 10 and 12 of a 21-day cycle
- A 38% objective response rate has been observed with mono-therapy in a heavily pretreated, serum RTX-neg DLBCL or mixed DLBCL/FL population; **of the subjects with RTX-neg DLBCL who received the MTD, 3 of 5 responded**
- The development of ADAs did not preclude benefit of MT-3724, consistent with what has been seen with other immunotoxins like denileukin diftitox and tagraxofusp-erzs
- The phase 2 portion of the study is ongoing, with sites open and actively enrolling globally
- MT-3724 is also being studied in combination with lenalidomide (NCT03645395) and gemcitabine/oxaliplatin (NCT03488251)

REFERENCES

1. Marshall MJE, et al. *Front Immunol.* 2017;8:1245.
2. Huang S, et al. *Blood Cancer J.* 2018;8:33.
3. Cheson BD, et al. *J Clin Oncol.* 2007;25:579-86.
4. Simon R. *Control Clin Trials.* 1989;10:1-10.

ACKNOWLEDGMENTS

The study was sponsored by Molecular Templates, Inc., and supported in part by a grant from Cancer Prevention & Research Initiative of Texas (CPRIT). Funding to support the development of this poster was provided by Molecular Templates to MedVal Scientific Information Services, LLC, Princeton, NJ.

DISCLOSURE

PAH: Consultancy: Juno, Sandoz, Karyopharm, Celgene, AstraZeneca; Research funding: Portola Pharmaceuticals, Molecular Templates, Incyte, Seattle Genetics, Novartis; DSMC and research funding: Janssen. **VM:** Principal investigator: Arensia EM; Employment: Institute of Oncology. **MZ:** Employment: ARENSIA Exploratory Medicine LLC. **SIP:** Research funding: Seattle Genetics, Teva, Takeda, and BMS; Member of speakers bureau: Gilead and Seattle Genetics; Member of advisory committee: Rafael Pharmaceuticals, BMS, and Teva. **CB:** Employment: Molecular Templates. **KD:** Employment: Molecular Templates. **ETW:** Employment: Molecular Templates. **JPH:** Employment and Equity Ownership: Molecular Templates. **DOP:** Consultancy: Sandoz.

Presented at the 61st American Society of Hematology (ASH) Annual Meeting & Exposition, December 7-10, 2019, Orlando, FL, USA

The study was sponsored by Molecular Templates, Inc.