Combining MDSC immunophenotyping with the HyperVOX computational technique: a usable input into pattern recognition neural networks for early detection of stage I/II breast cancer (BCa)

George A. Dominguez[1*], John Roop[1], Alexander Polo[1], Anthony Campisi[1], Dmitry I. Gabrilovich[2], Amit Kumar[1]

[1]Anixa Biosciences, Inc., San Jose, CA; [2]The Wistar Institute, Philadelphia, PA

BACKGROUND AND PURPOSE

❖The immune system is intricately involved with tumor development, and in particular, myeloid-derived suppressor cells (MDSCs) have been shown to be key contributors in supporting tumor progression and escape and thought to be used as biomarkers.[1-4]

❖Flow cytometry-based immunophenotyping is widely incorporated into clinical trials to measure numerous immune cell populations, and with recent developments in flow and mass cytometry, more and more cell parameters can now be measured at one time.

❖Flow cytometry data is analyzed using manual gating, but this laborious manual operator-driven analysis is known to be inconsistent between operators, especially with high-dimensional panels.

❖With more usage of high-dimensional flow assays for immune monitoring, it has been difficult to use them for diagnostic use due to the inefficiency of manual gating.

❖Here, we implemented a new computational technique called 'hypervoxelation of cytometry events', or HyperVOX, allowing us to analyze immunophenotyping data using pattern recognition neural networks (PRNNs) to detect early breast cancer (stage I/II) in the absence of manual gating.

METHODS: PATIENT POPULATION AND CLINICAL CHARACTERISTICS

With IRB approval, peripheral blood was collected from 117 biopsy-confirmed BCa patients and 88 healthy donor female (HDF) controls (**Table 1**). Peripheral blood mononuclear cells (PBMCs) were isolated up to 36 hours post-collection. We used standard multiparametric flow cytometry techniques to measure 16 different immune cell populations, including MDSCs, using a BD LSRII flow cytometer (**Figure 1**). All samples were collected using the same flow cytometry protocols and compensation matrices.

Table 1. **Clinicopathological characteristics of BCa and HDF subjects.**

	BCa	HDF
Total	117	88
Age (years),		
Median ± SD	59 ± 11.9	42 ± 15.5
< 35	2 (1.7%)	32 (36.4%)
35 – 50	34 (29.1%)	20 (22.7%)
> 50	81 (69.2%)	36 (40.9%)
Clinical Stage,		
I/IA/IB	74 (63.3%)	
II/IIA/IIB	32 (27.3%)	
III/IIIA/IIIC	7 (6.0%)	
IV	2 (1.7%)	
Unknown	2 (1.7%)	
Biomarkers,		
ER⁺PR⁺HER2⁻	65 (55.6%)	
ER⁻PR⁻HER2⁻	12 (10.3%)	
ER⁺PR⁻HER2⁻	10 (8.5%)	
ER⁺PR⁺HER2⁺	6 (5.1%)	
ER⁻PR⁻HER2⁺	5 (4.3%)	
ER⁺PR⁻HER2⁺	3 (2.6%)	
ER⁻PR⁺HER2⁻	1 (0.9%)	
Unknown	15 (12.8%)	

Figure 1. **Gating strategy for (A) myeloid and (B) lymphoid cell panels. Representative flow cytometry plots showing the gating strategy used to identify myeloid and lymphoid cells in peripheral blood from HD and breast cancer patients. Manually gated populations percentages were determined as percent of live cells (DAPI⁻).**

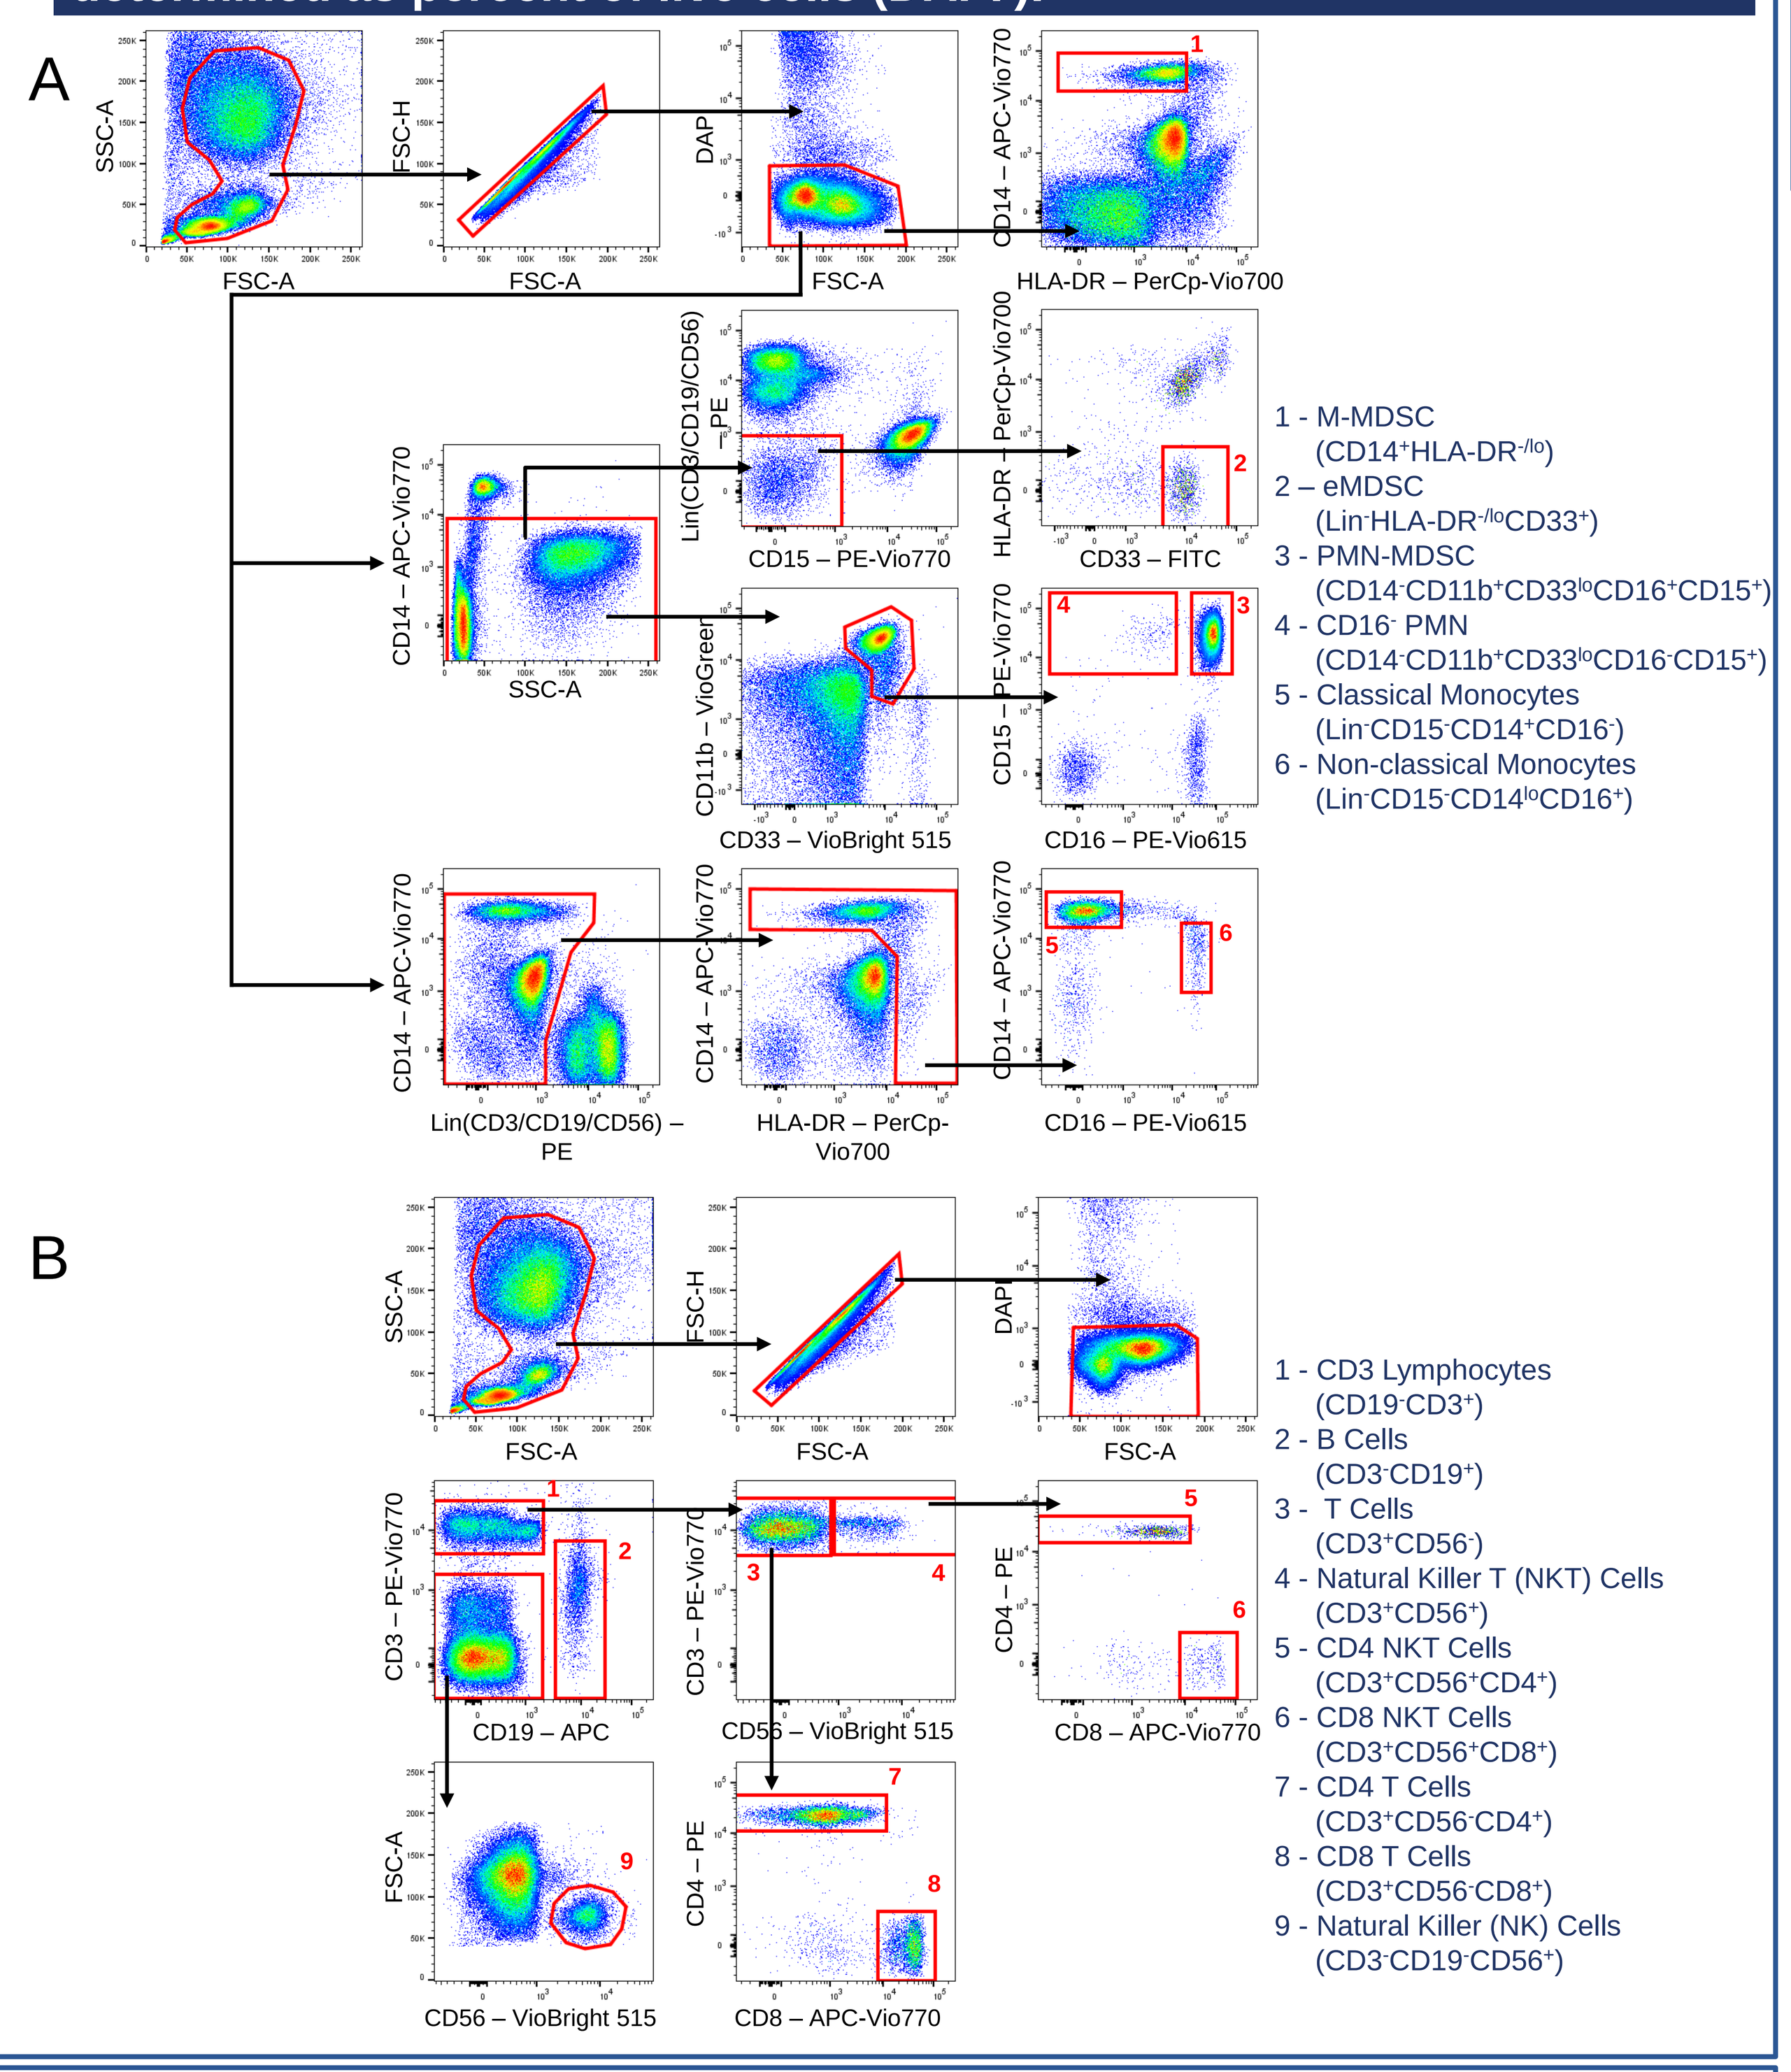
A
SSC-A
FSC-A
FSC-H
DAPI
CD14 – APC-Vio770
HLA-DR – PerCp-Vio700
Lin(CD3/CD19/CD56) – PE
CD15 – PE-Vio770
CD33 – FITC
SSC-A
CD11b – VioGreen
CD33 – VioBright 515
CD16 – PE-Vio615
Lin(CD3/CD19/CD56) – PE
HLA-DR – PerCp-Vio700
1 - M-MDSC (CD14⁺HLA-DR⁻/lo)
2 – eMDSC (Lin⁻HLA-DR⁻/loCD33⁺)
3 - PMN-MDSC (CD14⁻CD11b⁺CD33loCD16⁺CD15⁺)
4 - CD16⁻ PMN (CD14⁻CD11b⁺CD33loCD16⁻CD15⁺)
5 - Classical Monocytes (Lin⁻CD15⁻CD14⁺CD16⁻)
6 - Non-classical Monocytes (Lin⁻CD15⁻CD14loCD16⁺)
B
CD3 – PE-Vio770
CD19 – APC
CD56 – VioBright 515
CD4 – PE
CD8 – APC-Vio770
1 - CD3 Lymphocytes (CD19⁻CD3⁺)
2 - B Cells (CD3⁻CD19⁺)
3 - T Cells (CD3⁺CD56⁻)
4 - Natural Killer T (NKT) Cells (CD3⁺CD56⁺)
5 - CD4 NKT Cells (CD3⁺CD56⁺CD4⁺)
6 - CD8 NKT Cells (CD3⁺CD56⁺CD8⁺)
7 - CD4 T Cells (CD3⁺CD56⁻CD4⁺)
8 - CD8 T Cells (CD3⁺CD56⁻CD8⁺)
9 - Natural Killer (NK) Cells (CD3⁻CD19⁻CD56⁺)

METHODS: HYPERVOXELATION OF CYTOMETERY EVENTS (HyperVOX)

Hypervoxelated cytometry event counts were prepared using our novel procedure called 'hypervoxelation of cytometry events', or HyperVOX, wherein each channel of flow cytometry data was used as an axis in a multidimensional space. Each FCS file was imported into MATLAB, and each axis was then divided into four segments with each event being defined by its segment location on each axis. These regions are referred to as hypervoxels (**Figure 2**). Each hypervoxel is a feature common to all samples, and the count of events which fall in a particular hypervoxel for each sample is the distinct numerical value for each feature for that sample. This does not use conventional manual gating.

In this study, two separate panels were used each with a set number of flow parameters (*f*). The myeloid (*f* = 9) and lymphoid (*f* = 7) cell panels each contained a total number of hypervoxels calculated as 4^f. This resulted in a nine-dimensional array containing 4^9, or 262,144, hypervoxel locations for the myeloid cell panel, and a seven-dimensional array containing 4^7, or 16,384, hypervoxels locations for the lymphoid cell panel. Each location in the nine- and seven-dimensional arrays represented a position in the flow cytometry hyperspace and contained the number of events indexed to that location. A count was then made of the number of events falling within each hypervoxel - the common feature between each sample.

Figure 2. **Illustration hypervoxel generation using the HyperVOX technique on flow cytometry data.**

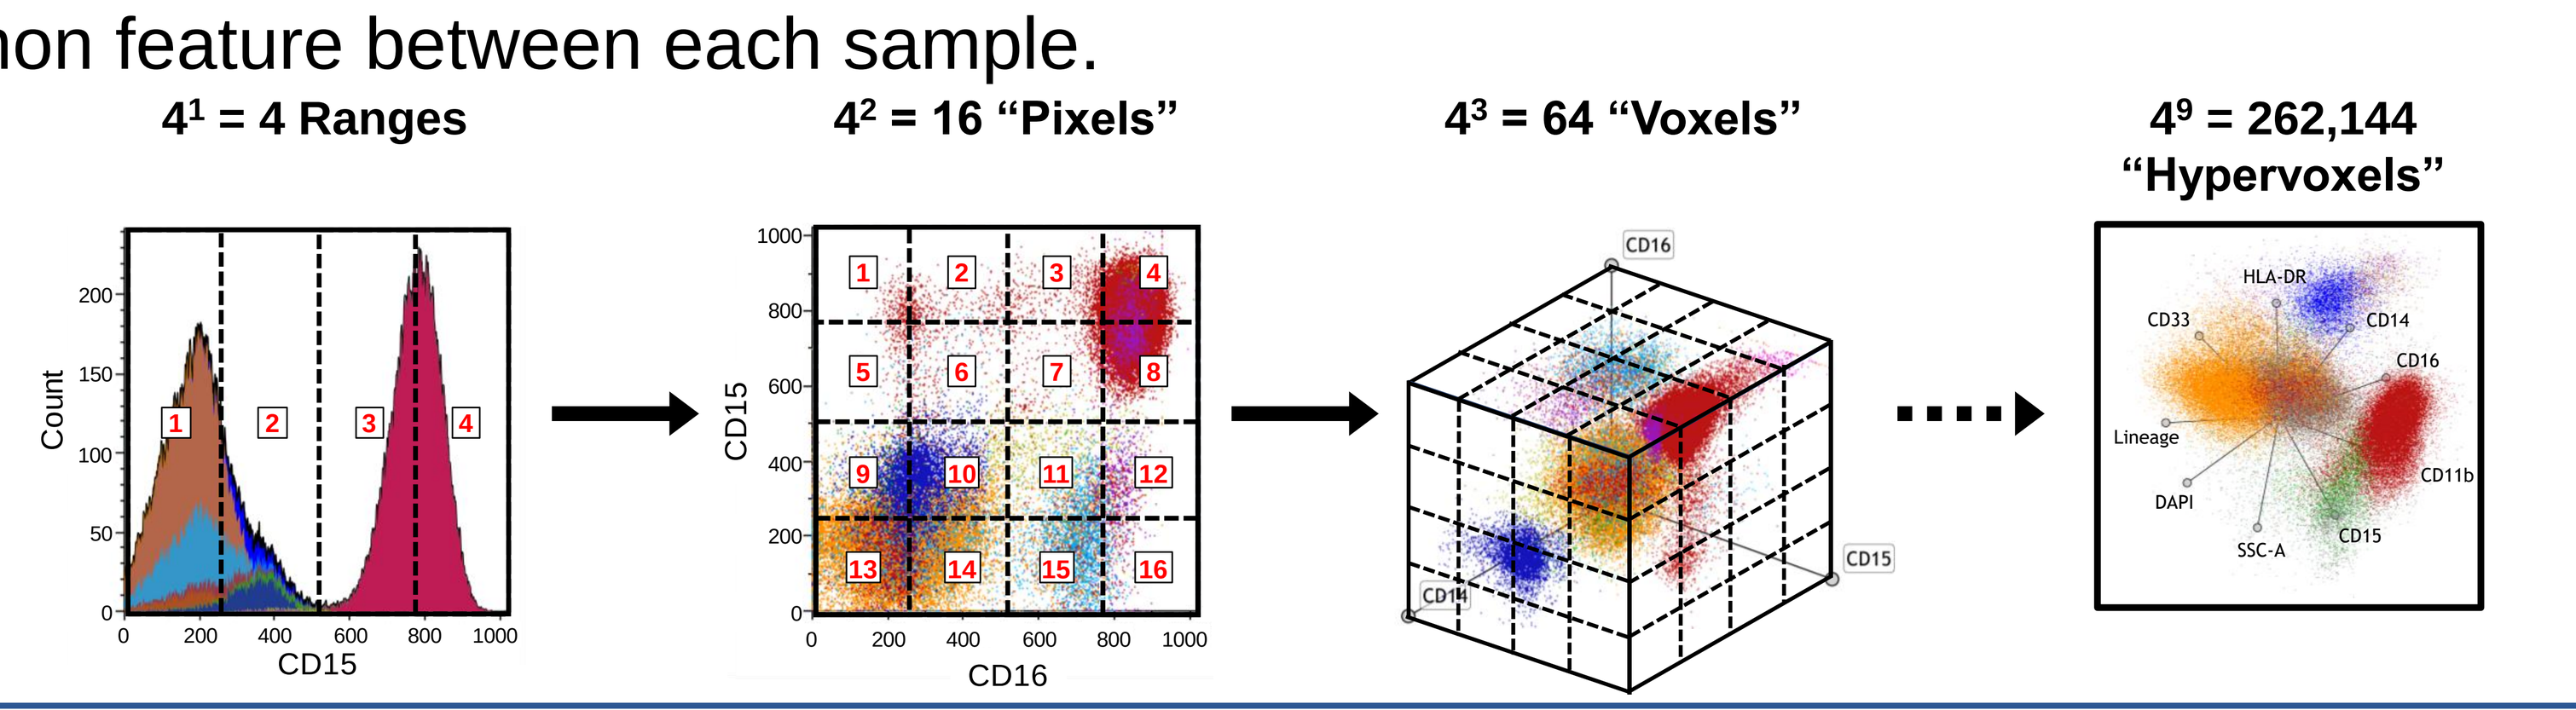
4¹ = 4 Ranges
4² = 16 "Pixels"
4³ = 64 "Voxels"
4⁹ = 262,144 "Hypervoxels"
Count
CD15
CD16
HLA-DR
CD33
CD14
CD11b
Lineage
DAPI
SSC-A

METHODS: PATTERN RECOGNITION NEURAL NETWORKS

For pattern recognition neural network training (PRNN), 500 different neural networks were trained using the three assembled datasets in 5 sets of 100 networks each (**Figure 3**). For each neural network trained, 80% of samples from each augmented class of patients were randomly selected for training. The remaining 20% from each class were used for network validation. The classes were balanced during both the training and validation processes. All training used scaled conjugate gradient backpropagation algorithms. For final evaluation, a separate holdout sample group, i.e. samples that were not used for PRNN training, of early stage BCa (I/II) were tested followed by subsequent sensitivity and specificity analysis.

Figure 3. **High level overview of pattern recognition neural network (PRNN) work flow. There are three input layers, one summing layer, and one output layer. The number of neurons used in each layer is listed beneath each neuron representation. w = weight, b = bias, and ∫ = activation function**

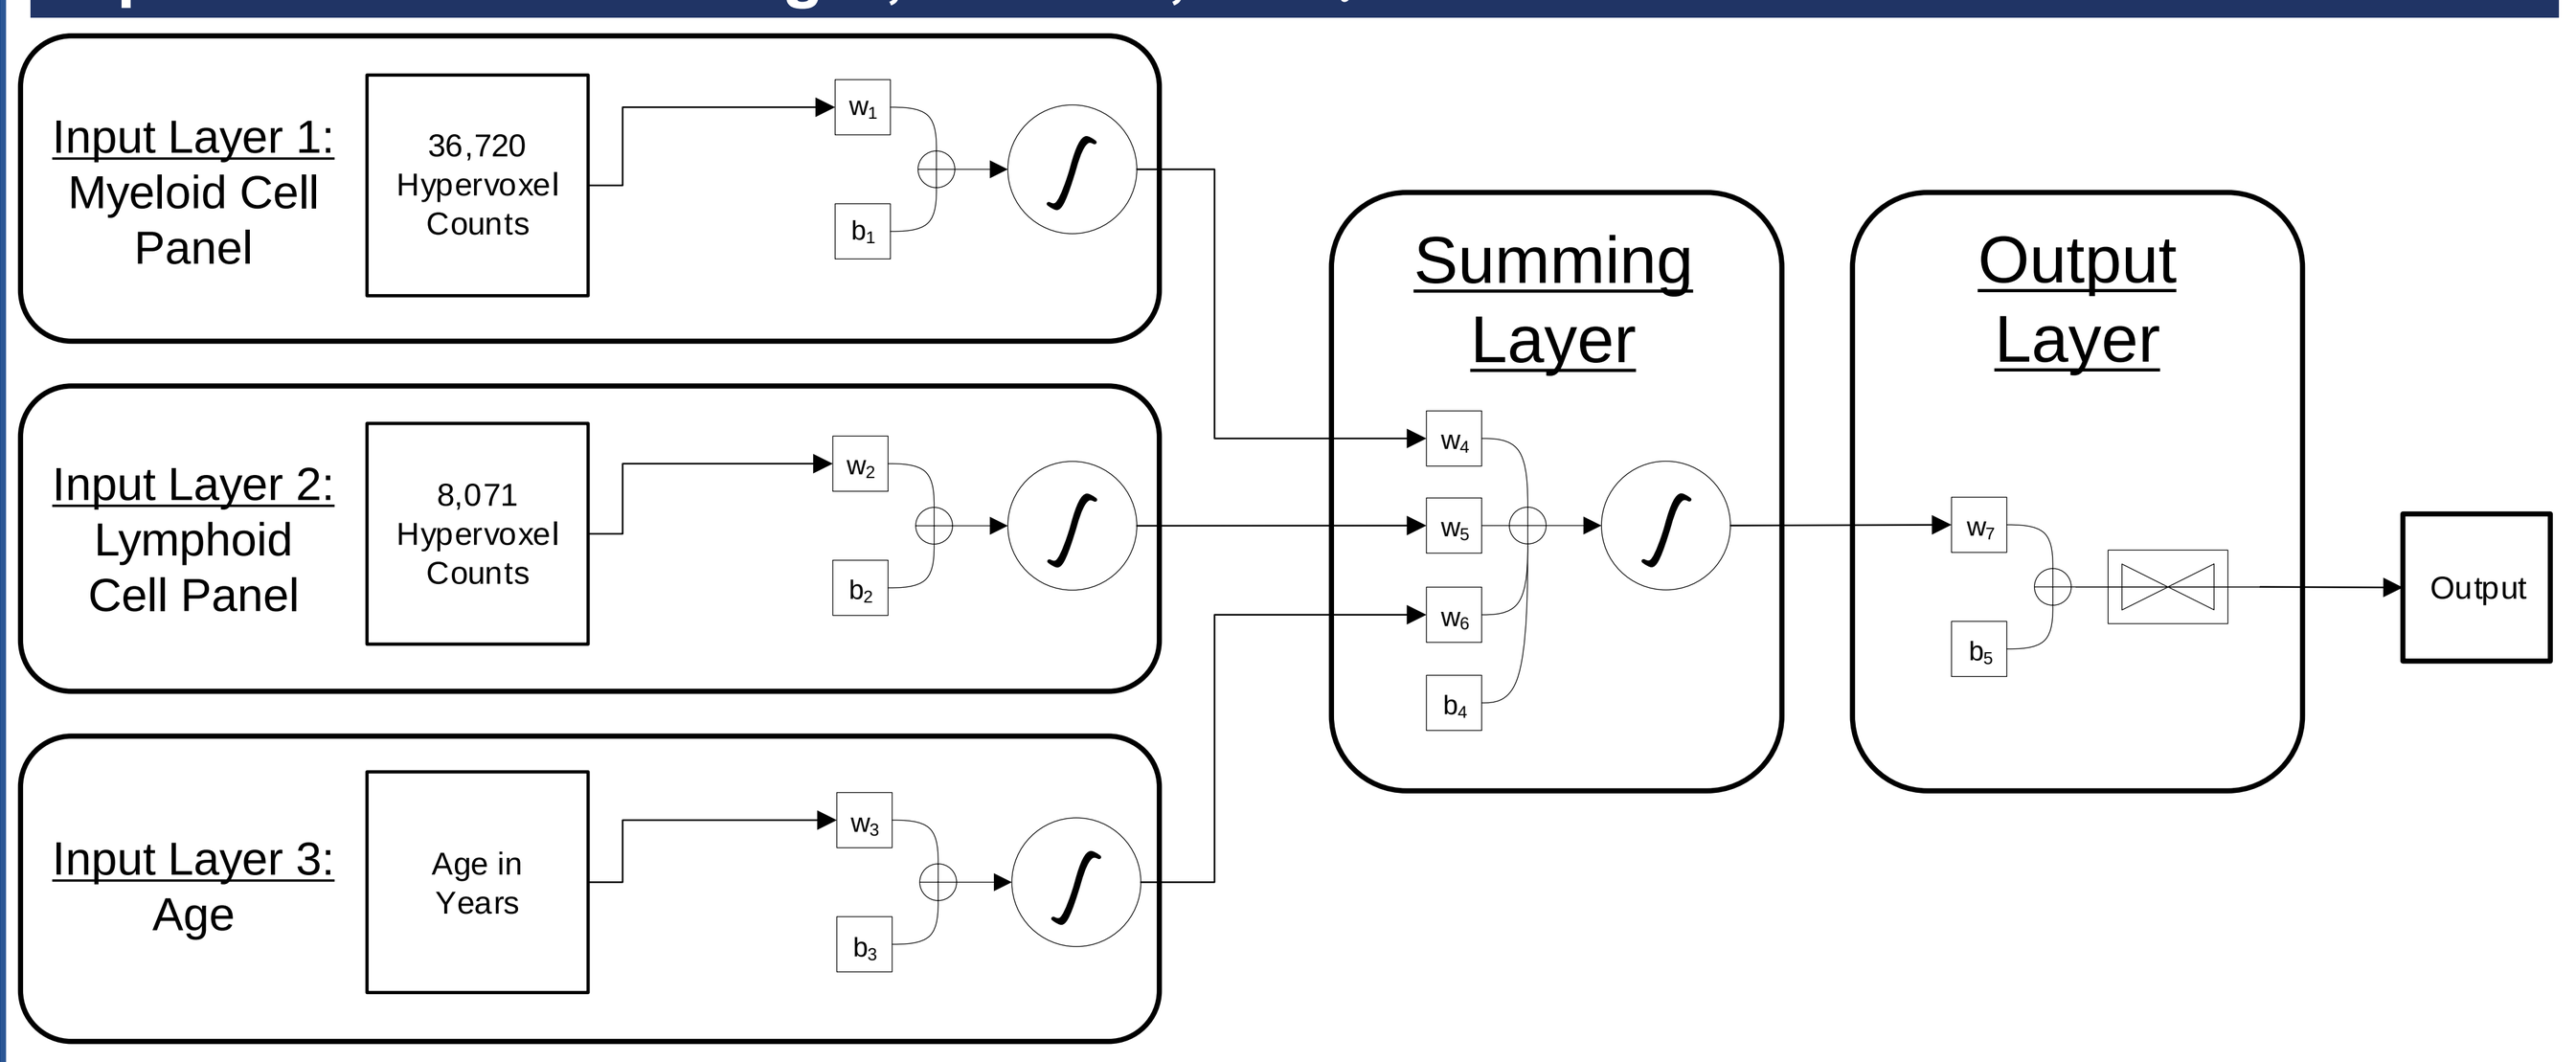
Input Layer 1: Myeloid Cell Panel
36,720 Hypervoxel Counts
Input Layer 2: Lymphoid Cell Panel
8,071 Hypervoxel Counts
Input Layer 3: Age
Age in Years
Summing Layer
Output Layer
Output

RESULTS: FLOW CYTOMETRY-BASED IMMUNOPHENOTYPING

✓Manual gating of MDSCs, myeloid, and lymphoid cell populations show trends with substantial overlap and are not specific enough to make accurate predictions (**Figure 4**).

✓There were no significant differences found in the levels of eMDSCs, PMN-MDSCs, and M-MDSCs between HDF, S1 BCa, or S2 BCa (**Figure 4A-C**).

✓There were significant increases in the CD14⁺ monocyte and classical monocyte populations (**Figure 4E and 4F**).

✓There were noted decreases in CD8⁺ T cell, NKT cell, CD4⁺ NKT, and CD8⁺ NKT cell populations (**Figure 4K, 4L, and 4N**).

Figure 4. **Comparisons of levels of different subsets of circulating myeloid and lymphoid cells between HDM and BCa patients. Peripheral blood from 88 HDF and 106 early stage BCa patients were stained for myeloid and lymphoid cell markers. Scatter plots show the median of calculated percentages with interquartile ranges of (A) eMDSCs, (B) PMN-MDSCs, (C) M-MDSCs, (D) CD16⁻ PMN, (E) CD14+ monocytes, (F) classical monocytes, (G), non-classical monocytes, (H) CD3⁺ lymphocytes, (I) T cells, (J) CD4T cells, (K) CD8T cells, (L) natural killer T (NKT) cells, (M) CD4NKT cells, (N) CD8NKT cells, (O) NK cells, and (P) B cells. * < 0.05, ** < 0.005, *** < 0.0005, and **** < 0.0001**

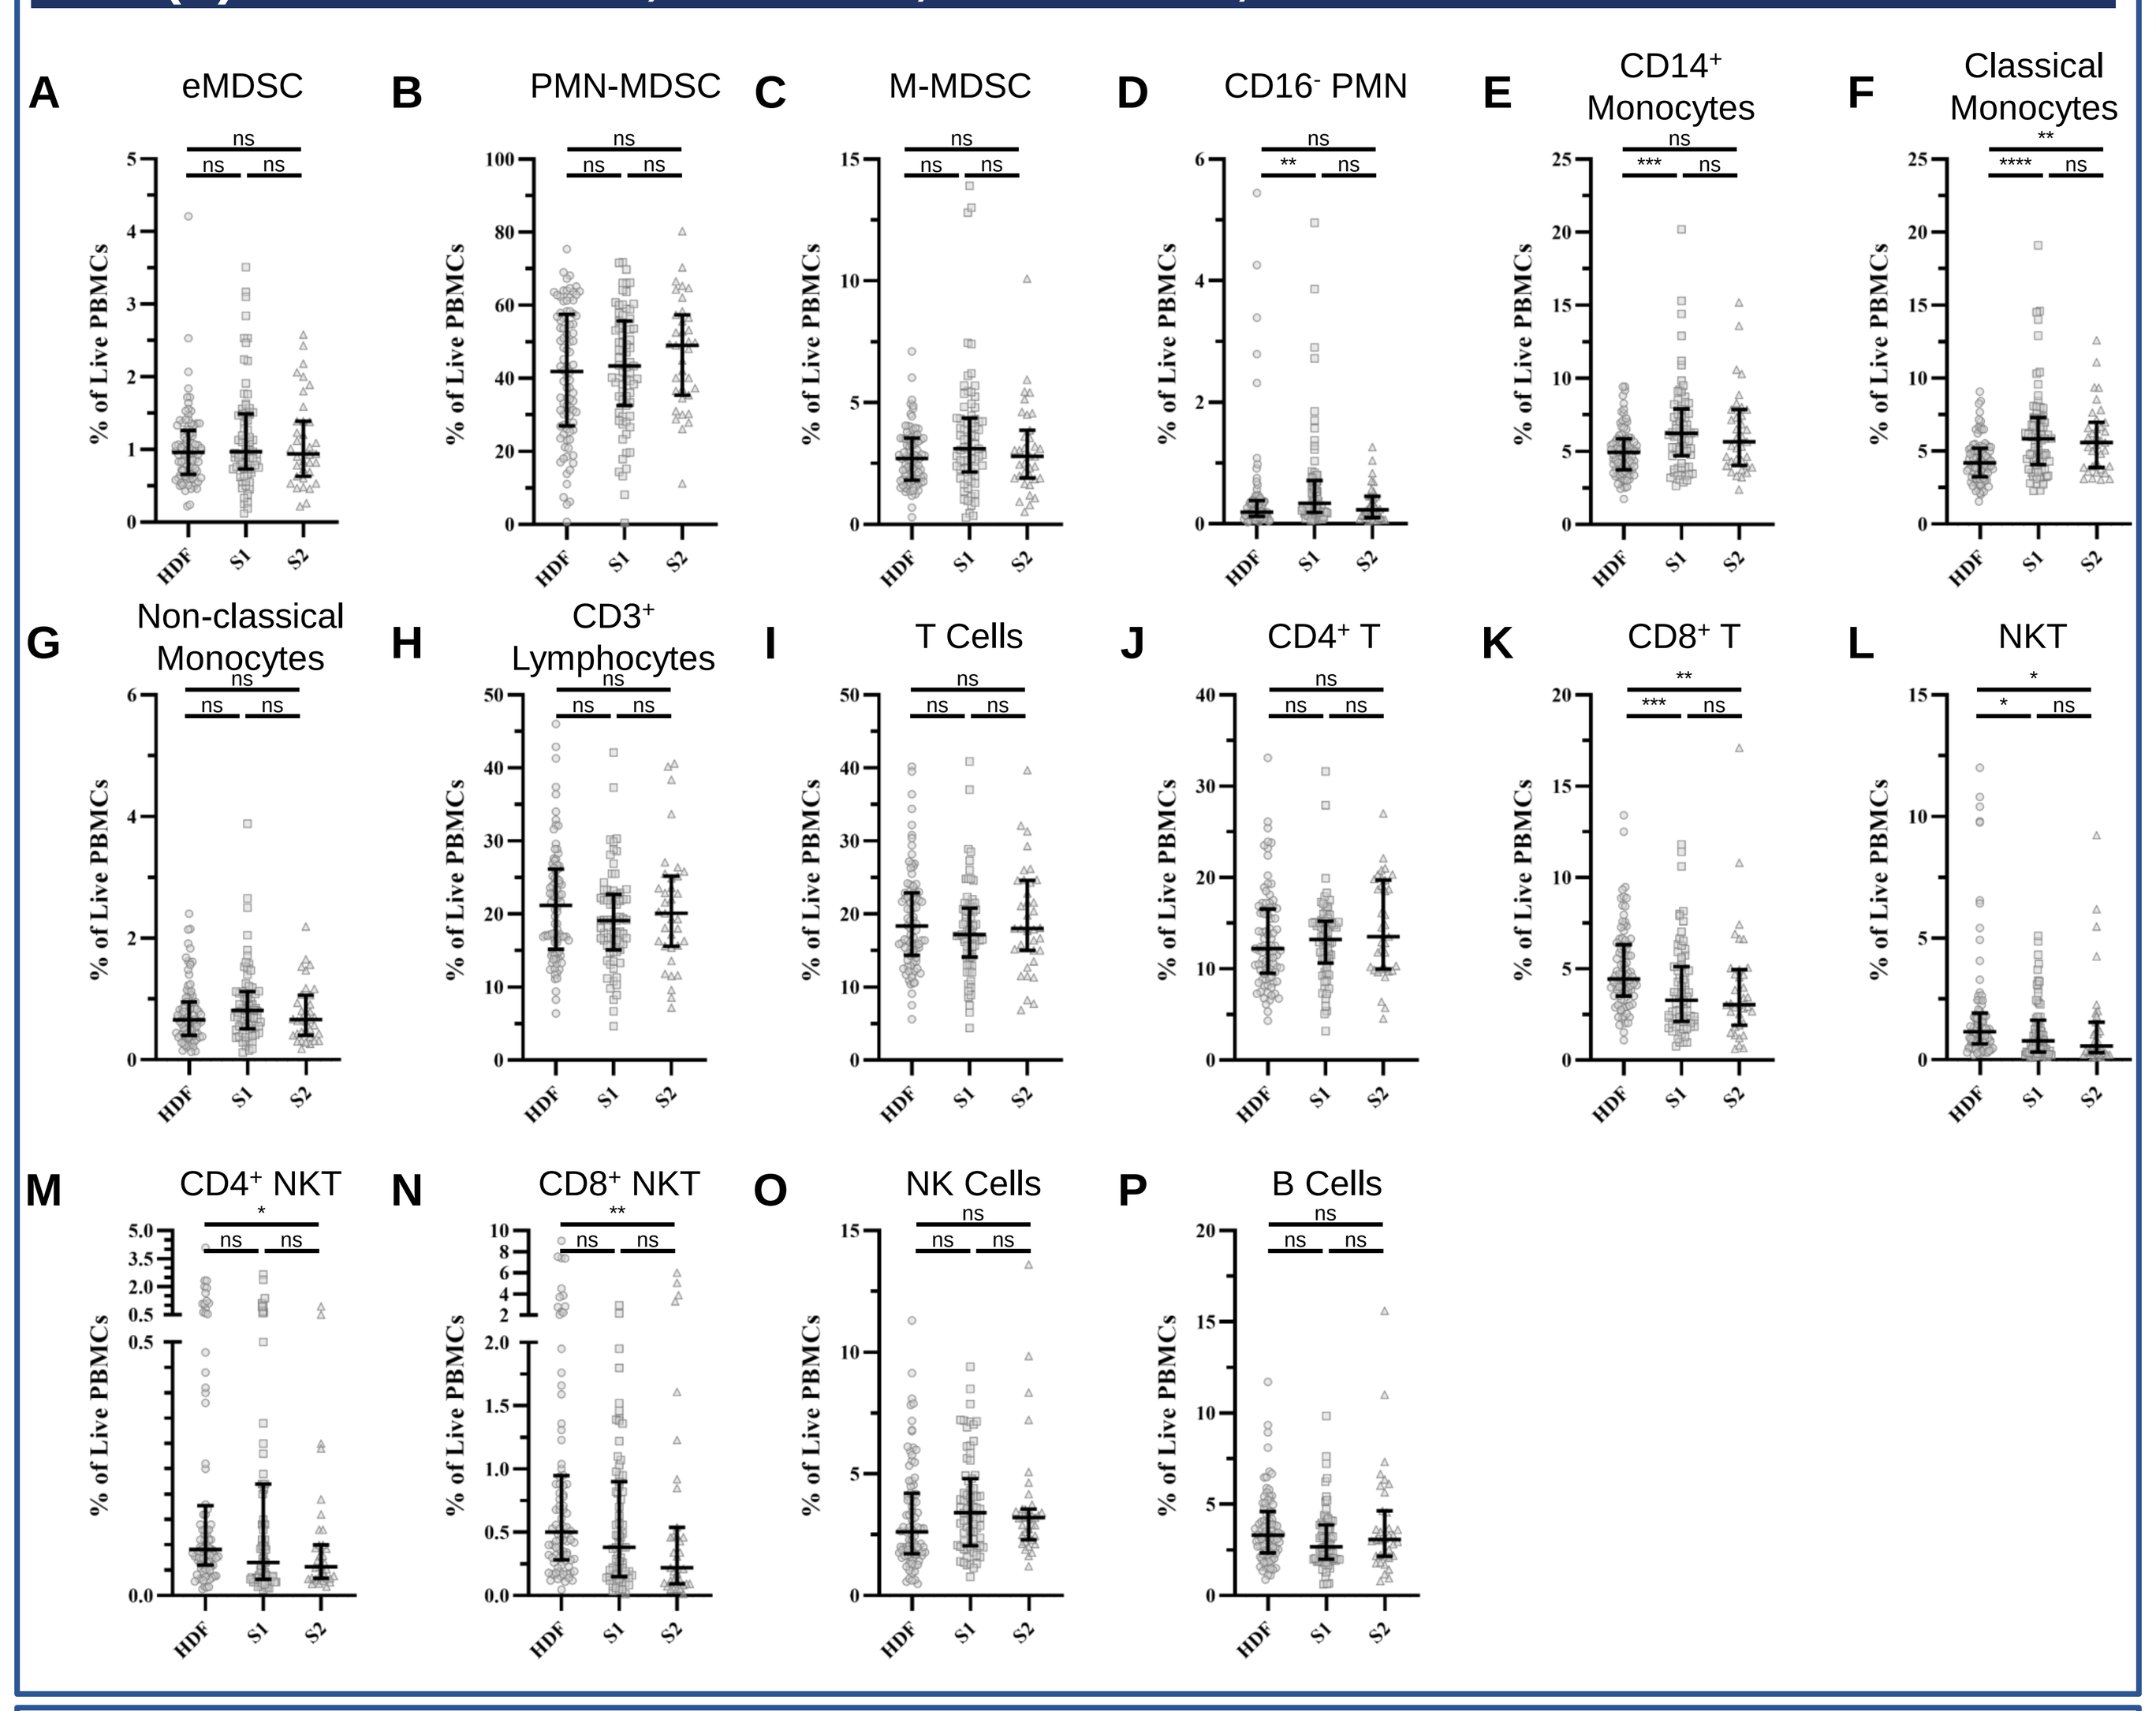
A eMDSC
B PMN-MDSC
C M-MDSC
D CD16⁻ PMN
E CD14⁺ Monocytes
F Classical Monocytes
G Non-classical Monocytes
H CD3⁺ Lymphocytes
I T Cells
J CD4⁺ T
K CD8⁺ T
L NKT
M CD4⁺ NKT
N CD8⁺ NKT
O NK Cells
P B Cells
% of Live PBMCs
HDF
S1
S2

STATISTICAL ANALYSIS

Manually gated population values were plotted as individual values with bars representing median and interquartile ranges. Statistical analysis was performed using non-parametric two-tailed Mann-Whitney U-test. All receiver operating characteristic (ROC) curves, area under the curve (AUC), 95% confidence intervals, and P-values were calculated using GraphPad Prism 7 or MATLAB software. Significance was determined at *P < 0.05*.

RESULTS: PRNN ANALYSIS TO DETECT BCa

✓The PRNN classifier was trained and validated using 69 HDF and 75 BCa patient samples.

✓A holdout sample dataset with 19 HDF and 42 stage I and II BCa patient samples was used to evaluate final PRNN classifier performance.

✓The PRNN classified 17 out of 19 HDF and 38 out of 42 BCa samples correctly resulting in a sensitivity of 90.48%, specificity of 89.47%, positive predictive value (PPV) of 95.00%, negative predictive value (NPV) of 80.95%, and accuracy of 90.16% **(TABLE 2)**.

✓The AUC was 0.9749 – far greater than using manually gated MDSC values (**Figure 5**).

✓Upon staging stratification, the PRNN classified 30 out of 30 stage I and 8 out of 12 stage II BCa samples correctly.

Table 2. **Sensitivity and specificity results of flow cytometry data for early breast cancer detection using HyperVOX and PRNN analysis.**

Statistic	Value	95% CI Interval
Sensitivity	90.48	77.38 – 97.34
Specificity	89.47	66.86 – 98.70
PPV	95.00	83.61 – 98.61
NPV	80.95	62.29 – 91.62
Accuracy	90.16	79.81 – 96.30
AUROC	0.9749	0.9391 – 1

Figure 5. **ROC curve generated from hypervoxelated flow cytometry data of holdout samples analyzed using PRNN analysis of patients with BCa versus HDF.**

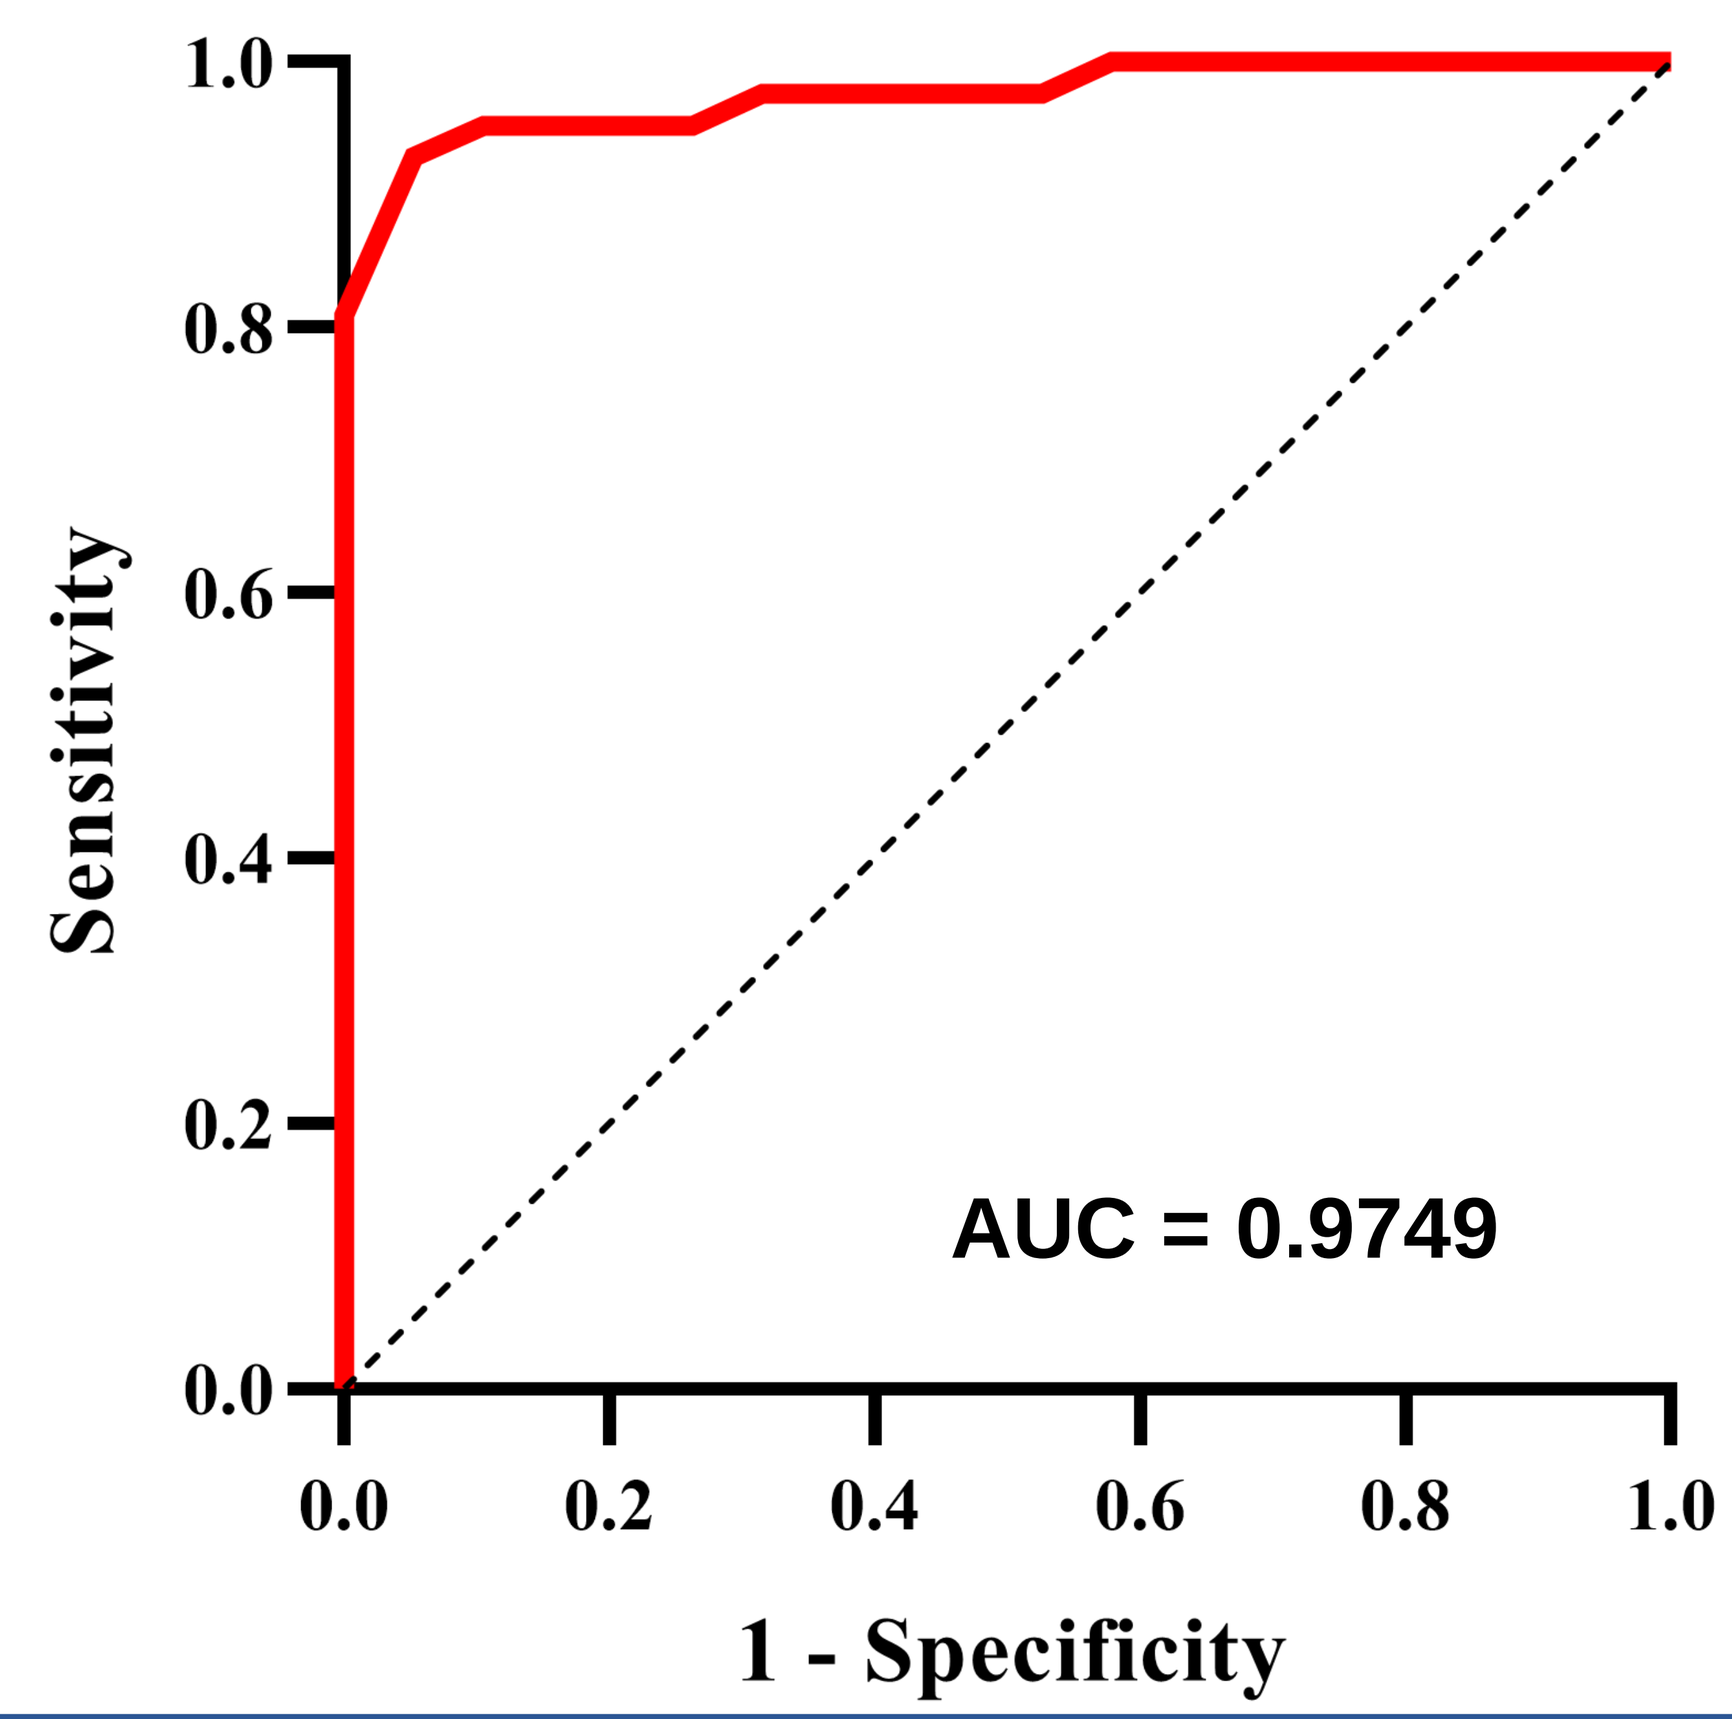
Sensitivity
1 - Specificity
AUC = 0.9749

CONCLUSIONS

➢To our knowledge, this is the first application for flow cytometry event data used to directly train pattern recognition neural networks (PRNNs) to detect early stage breast cancer based upon the immunophenotyping of MDSC, myeloid, and lymphoid cell populations.

➢The use of PRNN to analyze flow cytometry data with HyperVOX demonstrated high levels of accuracy that manual gating, even with numerous ML analysis methods, failed to achieve.

➢We expect classification to improve as more samples are included in the training sets and additional immune cell populations are included.

➢In a clinical setting, this type of assay could aid in the clinician's ability to better recommend breast biopsies and improve breast cancer detection non-invasively in conjunction with current detection modalities.

➢Furthermore, we believe that this technology could be used for other types of binary classifications, such as predicting patient responses to immunotherapies and/or monitoring for the recurrence of tumors.

REFERENCES

1. Kumar et al. *Trends Immunol*. 2016; 37:208-220.
2. Marvel et al. *J Clin Invest.* 2015; 125:3356-3364.
3. Elliott et al. *Front Immunol.* 2017; 8:86.
4. Okla et al. *Crit Rev Clin Lab Sci.* 2018.

ACKNOWLEDGEMENTS

The authors would like to show our greatest appreciation to the patients and their families for their participation as well as all clinicians and study staff.

Contact information: george@anixa.com

Presented at Cell Symposia: Hallmarks of Cancer; November, 17-19 2019; Seattle, Washington, USA