Targeting of misfolded, pathogenic TDP-43 with rationally designed antibodies

Neil Cashman[1,2,3], Sarah Louadi[1,2] , Beibei Zhao[1,2], Andrei Roman[1,2], Ebrima Gibbs[1,2], Anke Dijkstra[4], Johanne Kaplan[3]
[1]Djavad Mowafaghian Centre for Brain Health, [2]University of British Columbia, Vancouver BC, [3]ProMIS Neurosciences, Toronto ON;
[4]Amsterdam University Medical Center, Amsterdam, Netherlands

INTRODUCTION

Background

- Misfolded protein aggregates of TDP43 are a pathological hallmark of ALS and FTLD[1]
- The stereotypical distribution of TDP43 pathology in ALS and FTD shows a spreading pattern consistent with progressive dissemination from neuron to neuron[2,3]
- Misfolded aggregates of TDP43 are toxic to neural cells[4,5]
- Prion-like cell to cell propagation of TDP43 aggregates has been demonstrated in cell culture[5,6] and animal models[7]
- We have previously reported that pathogenic TDP43 induces misfolding of SOD1[8] and we recently determined that a tryptophan (Trp68) in the TDP-43 N-terminal domain (NTD) participates in this cross-seeding despite being inaccessible in the natively folded NTD[9]

Induction of SOD1 mis-folding by pathogenic TDP43[8]

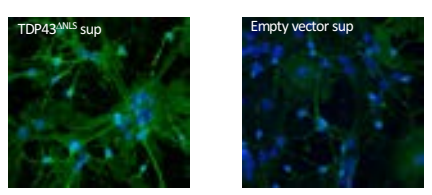
TDP43$^{\Delta NLS}$ sup
Empty vector sup

Mis-folded SOD1 (green) in primary spinal cord cultures exposed to supernatant from HEK293 cells transduced with TDP43^{DNLS} (misfolded) but not empty vector

Goal

Generate antibodies selective for misfolded disease-associated TDP-43 through immunization of rabbits with an unfolded NTD linear epitope including Trp68 (anti-NTD).

METHODS

Surface Plasmon Resonance:
•TDP43 NTD epitope-BSA conjugate was immobilized on sensor chips at a very low density of ~50 Rus. Rabbit mAbs were diluted 4-fold (31.25nM, 7.81nM, 1.95nM, 0.98nM, 0.24nM) and injected over the surface. The binding curves were fitted to a Langmuir 1:1 interaction model

Purification of TDP43 NTD
•Plasmid for purification of N-terminal domain of TDP-43 (residues 1-80) was generously provided by Nicolas L. Fawzi. Protein was expressed in Escherichia coli and purified as described previously[10]. Purified TDP-43 NTD was dialyzed against PBS, concentrated to 0.5 mg/ml, and and stored at −80 °C.

Western blot: SDS and Native gels
•Native-PAGE and SDS-PAGE were carried out using the Novex Bis-Tris system according to the manufacturer's specifications, then proteins were blotted onto PVDF membranes (Thermo Fisher Scientific, USA) . Blots were blocked in 5% milk in TBST, and then incubated with the anti-NTD Ab overnight at 4 °C followed with incubation with donkey anti-Rabbit IgG HRP-labelled secondary antibodies (GE Healthcare Life Sciences, USA). The SuperSignal West Femto (Thermo Scientific, USA) substrate was used according to the manufacturer's instructions.

Size Exclusion Chromatography
•Analytical gel-filtration of TDP43 NTD forms was performed using high performance liquid chromatography instrument on a Superdex 75 (10/300) HPLC column (GE Healthcare Life Sciences, USA). TDP-43 NTD (0.5 mg/ml) was denatured by incubation in 6 M Guanidine-HCl.

Immunocytochemistry:
•HEK293FT cells were labeled using a chicken anti-HA (abcam, USA), anti-pan-TDP-43 (Proteintech, USA) , and anti-NTD visualize TDP-43, and Hoechst 33342 for nuclear staining.
•For the visualization of stress granules with an anti-G3BP1 antibody (abcam, USA), cells were exposed to 1mM Sodium Arsenite for 60min before staining.

Immunohistochemistry:
•Cervical spinal cord sections from ALS and brain sections from FTLD donors were formalin-fixed and paraffin embedded, then stained with anti-NTD

Rabbit Ab raised against NTD epitope of misfolded TDP-43 recognize denatured but not natively folded TDP-43 N-terminus domain

Anti-NTD Abs display picomolar affinity for their cognate peptide epitope as assessed by surface plasmon resonance (SPR)

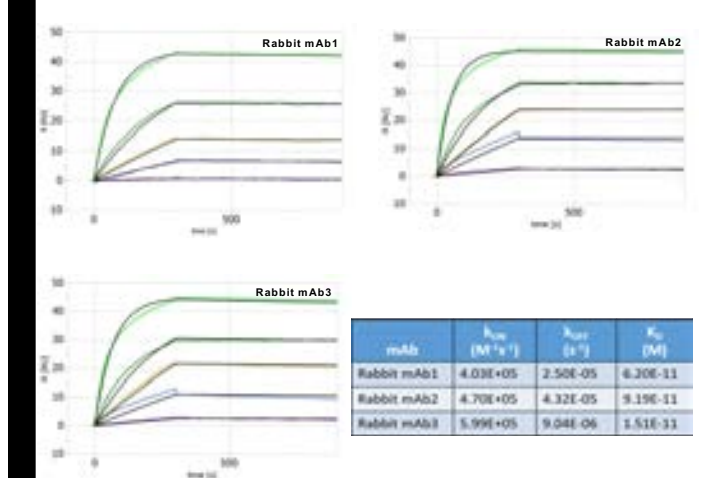
Rabbit mAb1
Rabbit mAb2
Rabbit mAb3

mAb	k_{on} ($M^{-1}s^{-1}$)	k_{off} (s^{-1})	K_D (M)
Rabbit mAb1	4.03E+05	2.50E-05	6.20E-11
Rabbit mAb2	4.70E+05	4.32E-05	9.19E-11
Rabbit mAb3	5.99E+05	9.04E-06	1.51E-11

Anti-NTD Ab reacts with denatured TDP-43 (SDS gel) but not native TDP-43

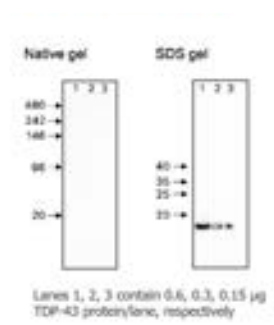
Native gel
SDS gel

Lanes 1, 2, 3 contain 0.6, 0.3, 0.15 µg TDP-43 protein/lane, respectively

Size exclusion chromatography confirms integrity of TDP-43 protein

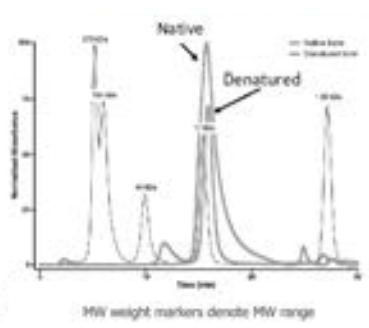
Native
Denatured

MW weight markers denote MW range

NTD epitope of misfolded TDP-43 is recognized by anti-NTD in brain tissue from FTLD patients with pathology subtypes A,B,C,E and in spinal cord from ALS patients

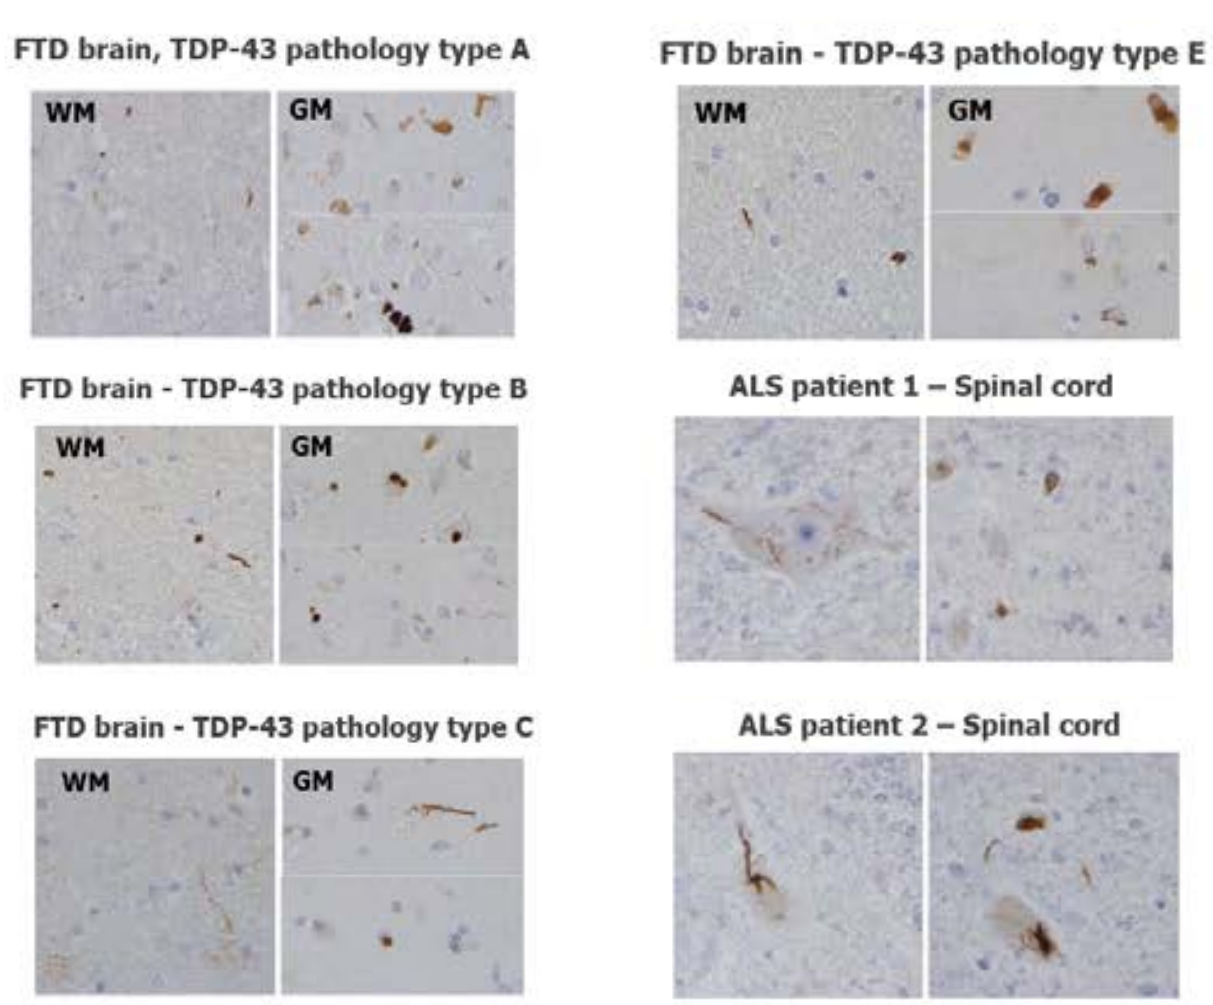
FTD brain, TDP-43 pathology type A
WM
GM
FTD brain - TDP-43 pathology type E
WM
GM
FTD brain - TDP-43 pathology type B
WM
GM
ALS patient 1 – Spinal cord
FTD brain - TDP-43 pathology type C
WM
GM
ALS patient 2 – Spinal cord

WM = White matter GM = Grey matter

Abs to misfolded TDP-43 NTD react with mislocalized, aggregated ΔNLS-TDP-43 and this immunoreactivity is abrogated by mutation of Trp68 to Ser68

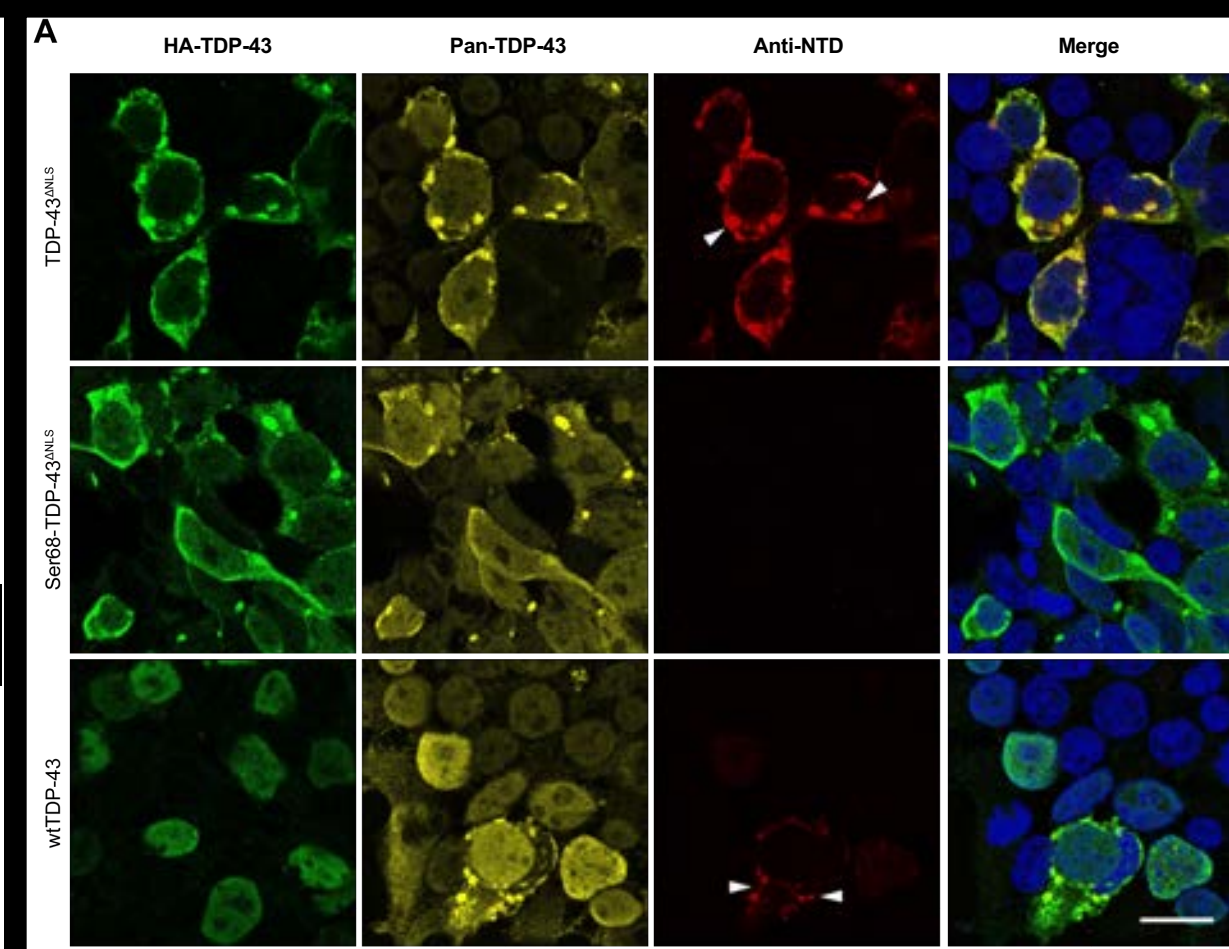
A
HA-TDP-43
Pan-TDP-43
Anti-NTD
Merge
TDP-43$^{\Delta NLS}$
Ser68-TDP-43$^{\Delta NLS}$
wtTDP-43

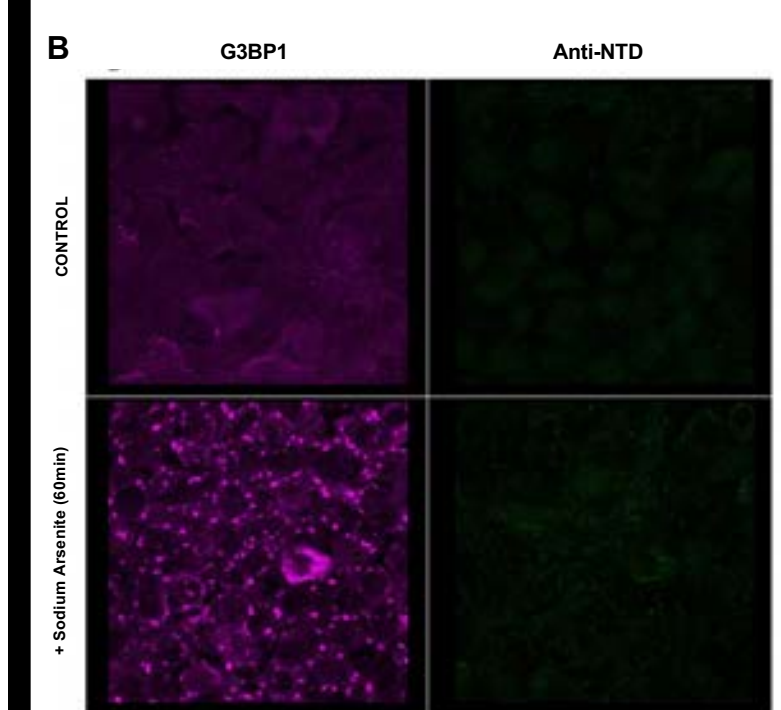
B
G3BP1
Anti-NTD
CONTROL
+ Sodium Arsenite (60min)

A. Anti-NTD (red) was tested for reactivity and specificity in cells transfected with different TDP-43 constructs. A pan-TDP-43 antibody against the C-terminal domain and an anti-HA-tag antibody were used to test co-localization with TDP-43 (yellow) and over-expressed TDP-43 (green) respectively. Anti-NTD specifically recognizes mislocalized cytoplasmic TDP-43 aggregates in TDP-43$^{\Delta NLS}$-transfected cells but not the ones where Trp68 is mutated (Ser68 TDP-43$^{\Delta NLS}$) nor does it recognize nuclear TDP-43. The composite images are a merge between nuclear, anti-NTD, and anti-HA-tag staining. This set of data suggests that accessibility of the Trp68 surrounding sequence to anti-NTD is a consequence of the TDP-43-NTD loss-of-structure accompanying aggregation and/or misfolding of TDP-43 and that anti-NTD is specific for aggregated TDP43 molecules. Scale bar: 20µm.

B. Anti-NTD was tested against stress granules generated by exposure to Sodium Arsenite. While these granules are positive for the marker G3BP1 (magenta), they are not bound by anti-NTD (green). This suggests that this antibody recognizes aberrant cytoplasmic TDP-43 aggregates without binding stress granules.

CONCLUSION

- Immunization with a predicted NTD epitope of misfolded TDP-43 produced a family of antibodies sensitive to solvent exposure of NTD Trp68 (anti-NTD)
- Anti-NTD displayed high epitope binding affinity in the picomolar range
- It also showed reactivity and specificity for aberrant cytoplasmic TDP43 aggregates in a HEK293 cell model, and no reactivity with wild-type nuclear TDP43 -> preserves normal, essential TDP43 function
- The same antibody did not show binding to physiological stress granules -> preserves stress-protective function
- Staining of TDP43 in human FTD and ALS samples confirmed reactivity with pathological TDP43
- These antibodies may find utility in biomarker and immunotherapy applications for TDP-43 associated diseases

REFERENCES

1. Neumann M, Sampathu DM, Kwong LK, et al. Ubiquitinated TDP-43 in frontotemporal lobar degeneration and amyotrophic lateral sclerosis. *Science*. 2006;314(5796):130-133. doi:10.1126/science.1134108;

2. Braak H, Brettschneider J, Ludolph AC, Lee VM, Trojanowski JQ, Del Tredici K. Amyotrophic lateral sclerosis--a model of corticofugal axonal spread. *Nat Rev Neurol*. 2013;9(12):708-714. doi:10.1038/nrneurol.2013.221

3. Brettschneider J, Del Tredici K, Irwin DJ, et al. Sequential distribution of pTDP-43 pathology in behavioral variant frontotemporal dementia (bvFTD) [published correction appears in Acta Neuropathol. 2015 Jun;129(6):929. Lee, Edward B [added]]. *Acta Neuropathol*. 2014;127(3):423-439. doi:10.1007/s00401-013-1238-y

4. Wang W, Wang L, Lu J, et al. The inhibition of TDP-43 mitochondrial localization blocks its neuronal toxicity. *Nat Med*. 2016;22(8):869-878. doi:10.1038/nm.4130

5. Nonaka T, Masuda-Suzukake M, Arai T, et al. Prion-like properties of pathological TDP-43 aggregates from diseased brains. *Cell Rep*. 2013;4(1):124-134. doi:10.1016/j.celrep.2013.06.007

6. Feiler MS, Strobel B, Freischmidt A, et al. TDP-43 is intercellularly transmitted across axon terminals. *J Cell Biol*. 2015;211(4):897-911. doi:10.1083/jcb.201504057

7. Porta S, Xu Y, Restrepo CR, et al. Patient-derived frontotemporal lobar degeneration brain extracts induce formation and spreading of TDP-43 pathology in vivo. *Nat Commun*. 2018;9(1):4220. Published 2018 Oct 11. doi:10.1038/s41467-018-06548-9

8. Pokrishevsky E, Grad LI, Cashman NR. TDP-43 or FUS-induced misfolded human wild-type SOD1 can propagate intercellularly in a prion-like fashion. *Sci Rep*. 2016;6:22155. Published 2016 Mar 1. doi:10.1038/srep22155

9. Afroz T, Hock EM, Ernst P, et al. Functional and dynamic polymerization of the ALS-linked protein TDP-43 antagonizes its pathologic aggregation. *Nat Commun*. 2017;8(1):45. Published 2017 Jun 29. doi:10.1038/s41467-017-00062-0

10. Wang A, Conicella AE, Schmidt HB, et al. A single N-terminal phosphomimic disrupts TDP-43 polymerization, phase separation, and RNA splicing. *EMBO J*. 2018;37(5):e97452. doi:10.15252/embj.201797452

ACKNOWLEDGMENTS