EFFECTS OF NSI-189 PHOSPHATE, A NEUROGENIC COMPOUND, ON QUANTITATIVE ELECTROENCEPHALOGRAPHY (QEEG) IN PATIENTS WITH MAJOR DEPRESSIVE DISORDER (MDD) DURING A PHASE IB RANDOMIZED, DOUBLE-BLIND, PLACEBO-CONTROLLED, MULTIPLE-ASCENDING-DOSE STUDY

Brett A. English, Pharm.D.[1], Ph.D., Jack Johnstone, Ph.D.[2], Karl Johe, Ph.D.[3], Lev Gertsik, M.D.[4], Molly Sherman, B.S.[1], Maurizio Fava, M.D.[5], Bettina Hoeppner, Ph.D.[5], Martina Flynn[5], David Mischoulon, M.D., Ph.D. [5], Gustavo Kinrys, M.D.[5], Marlene Freeman, M.D.[5], and Larry Ereshefsky, Pharm.D., BCPP[1]

1. PAREXEL International, Glendale, CA.
2. Q-Metrix, Los Angeles, CA
3. Neuralstem, Inc., Germantown, MD
4. California Clinical Trials Medical Group, Glendale, CA
5. Harvard Medical School, Clinical Trials Network & Institute (CTNI), Massachuesetts General Hospital, Boston, MA

PAREXEL®
YOUR JOURNEY. OUR MISSION.™

BACKGROUND[1-8]

Major depressive disorder (MDD) is a common psychiatric disorder with an estimated prevalence of about 7% and results in significant disruption to occupational and social functioning. The pathophysiology of MDD is unknown, however preclinical and clinical data support the hypothesis that inhibition of neurogenesis of the hippocampus, by chronic exposure to stress hormones (i.e., cortisol), plays a significant role in the etiology of a number of psychiatric disorders and that reversal of impaired hippocampal neurogenesis may mediate the therapeutic effects of antidepressants. Antidepressants such as the selective serotonin reuptake inhibitors (SSRI) (i.e., citalopram) have been shown to increase hippocampal neurogenesis via actions on brain-derived neurotrophic factors (BDNF).

While few biomarkers of antidepressant treatment response exist, qEEG characterization of the posterior alpha rhythm (8-13 Hz) has been used as an index of cortical deactivation, where patients with MDD exhibit higher alpha power relative to controls. Laterality of alpha EEG has also been shown to change in response to acute doses of cortisol. Changes in posterior alpha frequency has been associated with treatment response to antidepressants such as SSRIs compared with non-responders.

NSI-189, a benzylpiperizine-aminopyridine, is a novel molecule developed by Neuralstem, Inc. for the treatment of MDD, based upon preclinical data demonstrating stimulation of neurogenesis of human hippocampus-derived neural stem cells *in vitro* and in mouse hippocampus *in vivo*. NSI-189 also exhibits preclinical antidepressant properties as demonstrated in the novelty suppressed feeding after 28-day oral dosing.

Previously reported, NS-189 demonstrated minimal improvement among the placebo-treated patients; at day 28, the efficacy measurements showed a clinically meaningful reduction in depressive and cognitive symptoms across all measures for the two lower doses (40 mg/day and 80 mg/day) but not for the highest dose (120 mg/day). These improvements appeared to persist over time during the follow-up for Montgomery-Asberg Depression Rating Scale (MADRS), the Symptoms of Depression Questionnaire (SDQ), and the Cognitive and Physical Functioning Questionnaire (CPFQ).

In a Ph1b, double-blind, placebo-controlled, multiple-ascending-dose study, patients with symptomatic MDD were randomized to receive NSI-189 40, 80, or 120 mg daily or placebo for 28 days. Assessment of alpha frequency using qEEG was used to characterize pharmacodynamic antidepressant effects of NSI-189 phosphate in patients with MDD.

METHODS

Study design

This is a Phase Ib, double-blind, randomized, placebo-controlled, multiple-dose study with 3 ascending cohorts. Twenty-four patients with MDD were recruited, with their diagnosis and illness severity confirmed through an independent, remote SAFER interview from the MGH CTNI raters. Each cohort also included at least 3 female subjects. Each patient underwent a Screening for eligibility (Day -37 to Day -6 or -3) and eligible patients were admitted into the unit on Day -5 to complete antidepressant washout and undergo baseline assessments. Patients were randomized (3:1) to receive NSI-189 phosphate or placebo for 28 days. Cohort 1 received NSI-189 (or placebo) 40 mg QD; Cohort 2, NSI-189 (or placebo) 40 mg BID; Cohort 3, NSI-189 (or placebo) 40 mg TID. During the 28-day, multiple-dose period, patients underwent traditional safety and pharmacokinetic assessments. Cognitive and behavioral assessments included the MGH CPFQ, CGI-S, C-SSRS, MADRS, and the SDQ.

In addition to the assessments above, several pharmacodynamic measures were obtained as exploratory biomarkers and included plasma BDNF, salivary and plasma cortisol and corticotropin-releasing factor (CRF) and qEEG measurements. Safety EEGs were performed at Screening and on Day 1, 14 and 28, at 1hr and 3hrs post-dose, bracketing the predicted Tmax of NSI-189 and were relative to the dosing frequency (i.e., Cohort 1 = after first dose; Cohort 2 = after 2nd dose, etc). Additional safety EEGs were performed on Day 3 and 7 for Cohorts 2 and 3. qEEG assessments to characterize antidepressant treatment response were performed 6hrs post-dose at Screening, and on Day 14 and 28 (Figure 1).

© 2014 PAREXEL International Corporation. All rights reserved.

Figure 1. Study timeline for safety and qEEG assessments

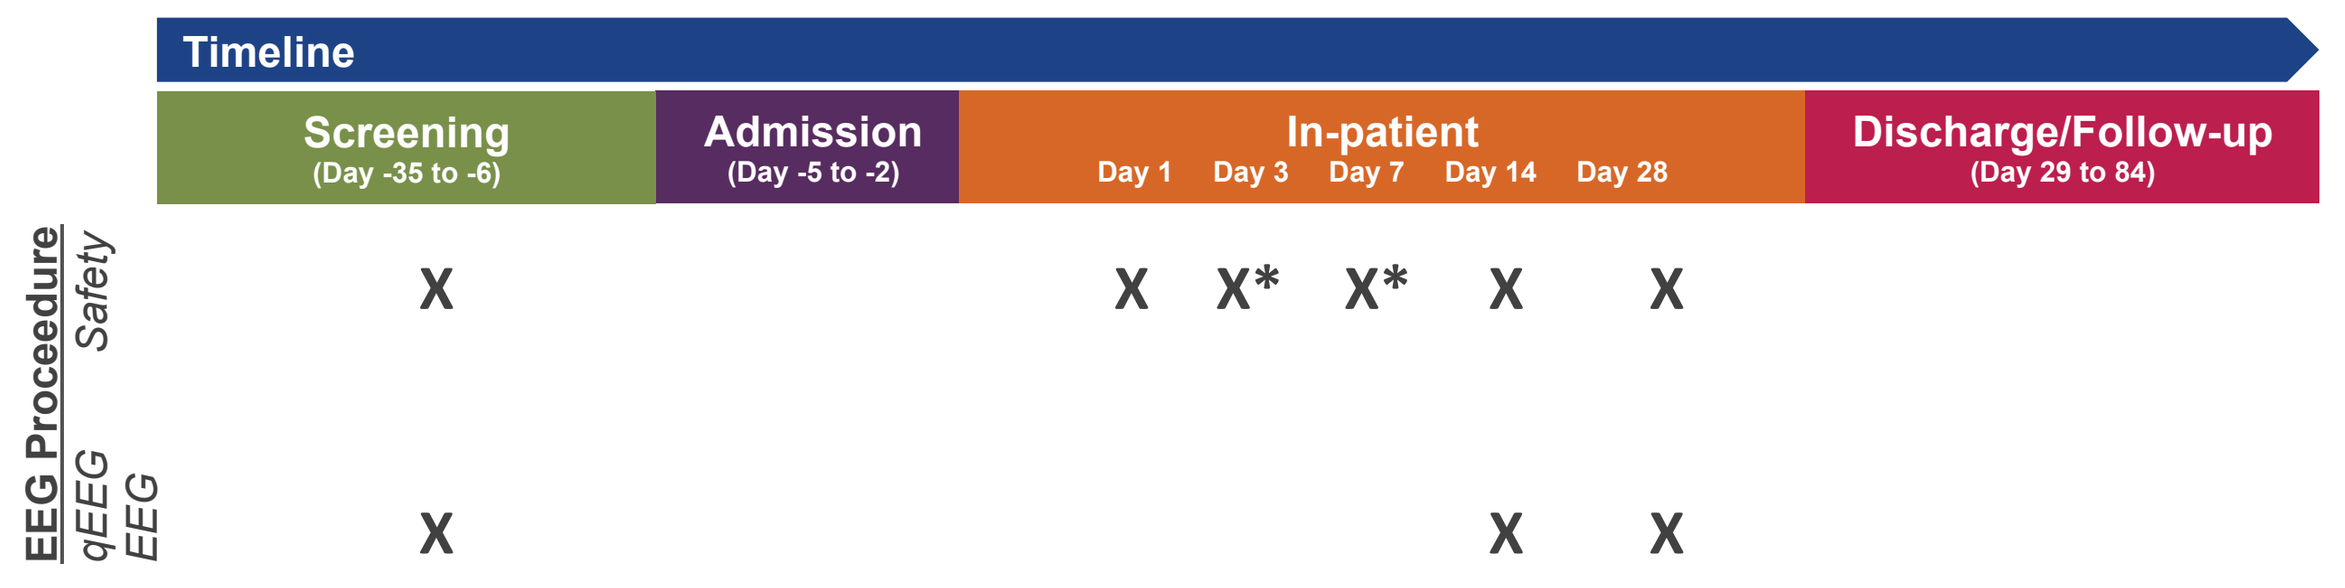
Timeline
Screening
(Day -35 to -6)
Admission
(Day -5 to -2)
In-patient
Day 1
Day 3
Day 7
Day 14
Day 28
Discharge/Follow-up
(Day 29 to 84)
EEG Proceedure
Safety
qEEG
EEG
X
X X* X* X X
X
X X

EEG procedures

Safety and quantitative EEG was recorded from 19 standard scalp locations, including EKG and eye-movement monitoring for 20-minutes. The 20 minute recording was performed at rest with eyes closed/eyes open and with hyperventilation and photic stimulation. All recordings were made with Cadwell Laboratories Easy II digital EEG instrumentation (Kennewick, WA) and evaluated by a trained clinical electrophysiologist for the identification of clinically significant findings at baseline and for subsequent recordings during dosing. qEEG was collected at Screening and Days 14 and 28. Safety EEG was collected at 1hr and 3hrs post-dose, relative to the dosing interval (ex: QD = after first dose; BID = after second dose, etc.).

Quantitative EEG analysis

Digital EEGs were reviewed to identify the presence of physiological and instrumentation artifact (i.e., eye blink, EMG activity, electrode movement, drowsiness) prior to qEEG recording. Artifacts were removed from EEG files manually by an experienced EEG technologist under the supervision of a trained clinical electrophysiologist. In some cases, artifacts are essentially continuous (i.e., EMG in frontal and temporal leads) and thus were not excluded from analyses. Extracerebral artifact is not considered to have a significant effect on the results of qEEG analyses as they do not contain analyses of frequencies in the EMG range. Typically 60-90 sec of EEG data are submitted to power spectral analyses using Brain Vision Analyzer software. These analyses generate an estimate of amplitude over specific frequency ranges: 4-6 Hz, 6-8 Hz, 8-20 Hz and 10-12 Hz for each of the 19 electrodes. In addition, EEG coherence measures, an index of phase consistency between electrode pairs, were computed over the same frequency ranges. Data were pooled (i.e., active total vs placebo total) and reported below.

RESULTS

Table 1. Patient demographics

		Placebo	40 mg QD	40 mg BID	40 mg TID	*P*
Ethnic origin, n (%)						0.604†
	Caucasian	4 (66.6)	3 (50.0)	2 (33.3)	2 (33.3)	
	African American	0 (0)	2 (33.3)	3 (50.0)	2 (33.3)	
	Hispanic	2 (33.3)	1 (16.6)	1 (16.6)	1 (16.6)	
	Asian	0 (0)	0 (0)	0 (0)	1 (16.6)	
Gender, n (%)						0.262†
	Male	5 (83.3)	2 (33.3)	3 (50.0)	2 (33.3)	
	Female	1 (16.6)	4 (66.6)	3 (50.0)	4 (66.6)	
Age (yr), mean (SD)		28.2 (4.75)	34.0 (3.50)	38.5 (12.94)	40.5 (9.92)	0.098*
MADRS total score at screening, mean (SD)		25.17 (2.93)	26.0 (3.35)	26.83 (2.64)	24.83 (2.56)	0.636*

SD = standard deviation

† Statistical method used, X^2 test.

* Statistical method used, 1-way analysis of variance (ANOVA).

Table 2. Clinically abnormal EEG findings*

Visit	Category*	40 mg QD NSI-189	40 mg QD PBO	40 mg BID NSI-189	40 mg BID PBO	40 mg TID NSI-189	40 mg TID PBO
Screening		6	2	7	3	6	2
	Normal, n (%)	6 (100)	2 (100)	5 (71.4)	3 (100)	4 (66.7)	1 (50)
	Abnormal, n (%)	0 (0)	0 (0)	2 (28.6)	0 (0)	2 (33.3)	1 (50)
Day 1		6	2	7	3	6	2
1hr post-dose	*Normal, n (%)*	6 (100)	2 (100)	6 (85.7)	2 (66.7)	5 (83.3)	2 (100)
	Abnormal, n (%)	0 (0)	0 (0)	1 (14.3)	1 (33.3)	0 (0)	0 (0)
3hr post-dose	*Normal, n (%)*	6 (100)	2 (100)	7 (100)	2 (66.7)	5 (83.3)	2 (100)
	Abnormal, n (%)	0 (0)	0 (0)	0 (0)	1 (33.3)	0 (0)	0 (0)
Day 3		6	2	6	3	6	2
1hr post-dose	*Normal, n (%)*	0 (0)	0 (0)	5 (83.3)	2 (66.7)	6 (100)	2 (100)
	Abnormal, n (%)	0 (0)	0 (0)	1 (16.7)	0 (0)	0 (0)	0 (0)
3hr post-dose	*Normal, n (%)*	0 (0)	0 (0)	6 (100)	2 (66.7)	6 (100)	2 (100)
	Abnormal, n (%)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)
Day 7		6	2	6	3	6	2
1hr post-dose	*Normal, n (%)*	0 (0)	0 (0)	5 (83.3)	2 (66.7)	6 (100)	2 (100)
	Abnormal, n (%)	0 (0)	0 (0)	1 (16.7)	1 (33.3)	0 (0)	0 (0)
3hr post-dose	*Normal, n (%)*	0 (0)	0 (0)	6 (100)	2 (66.7)	6 (100)	2 (100)
	Abnormal, n (%)	0 (0)	0 (0)	0 (0)	1 (33.3)	0 (0)	0 (0)
Day 14		6	2	6	2	6	2
	Normal, n (%)	5 (83.3)	2 (100)	5 (83.3)	2 (100)	6 (100)	1 (50)
	Abnormal, n (%)	1 (16.7)	0 (0)	1 (16.7)	0 (0)	0 (0)	1 (50)
Day 28		6	2	6	2	6	2
	Normal, n (%)	4 (66.7)	2 (100)	6 (100)	1 (50)	6 (100)	2 (100)
	Abnormal, n (%)	2 (33.3)	0 (0)	0 (0)	1 (50)	0 (0)	0 (0)

*EEG recordings were considered clinically mildly abnormal if they consisted of slowed or high-frequency activity regionally, but that were not considered epileptogenic by the clinical electrophysiologist.

Figure 2. High-frequency alpha at Day 28

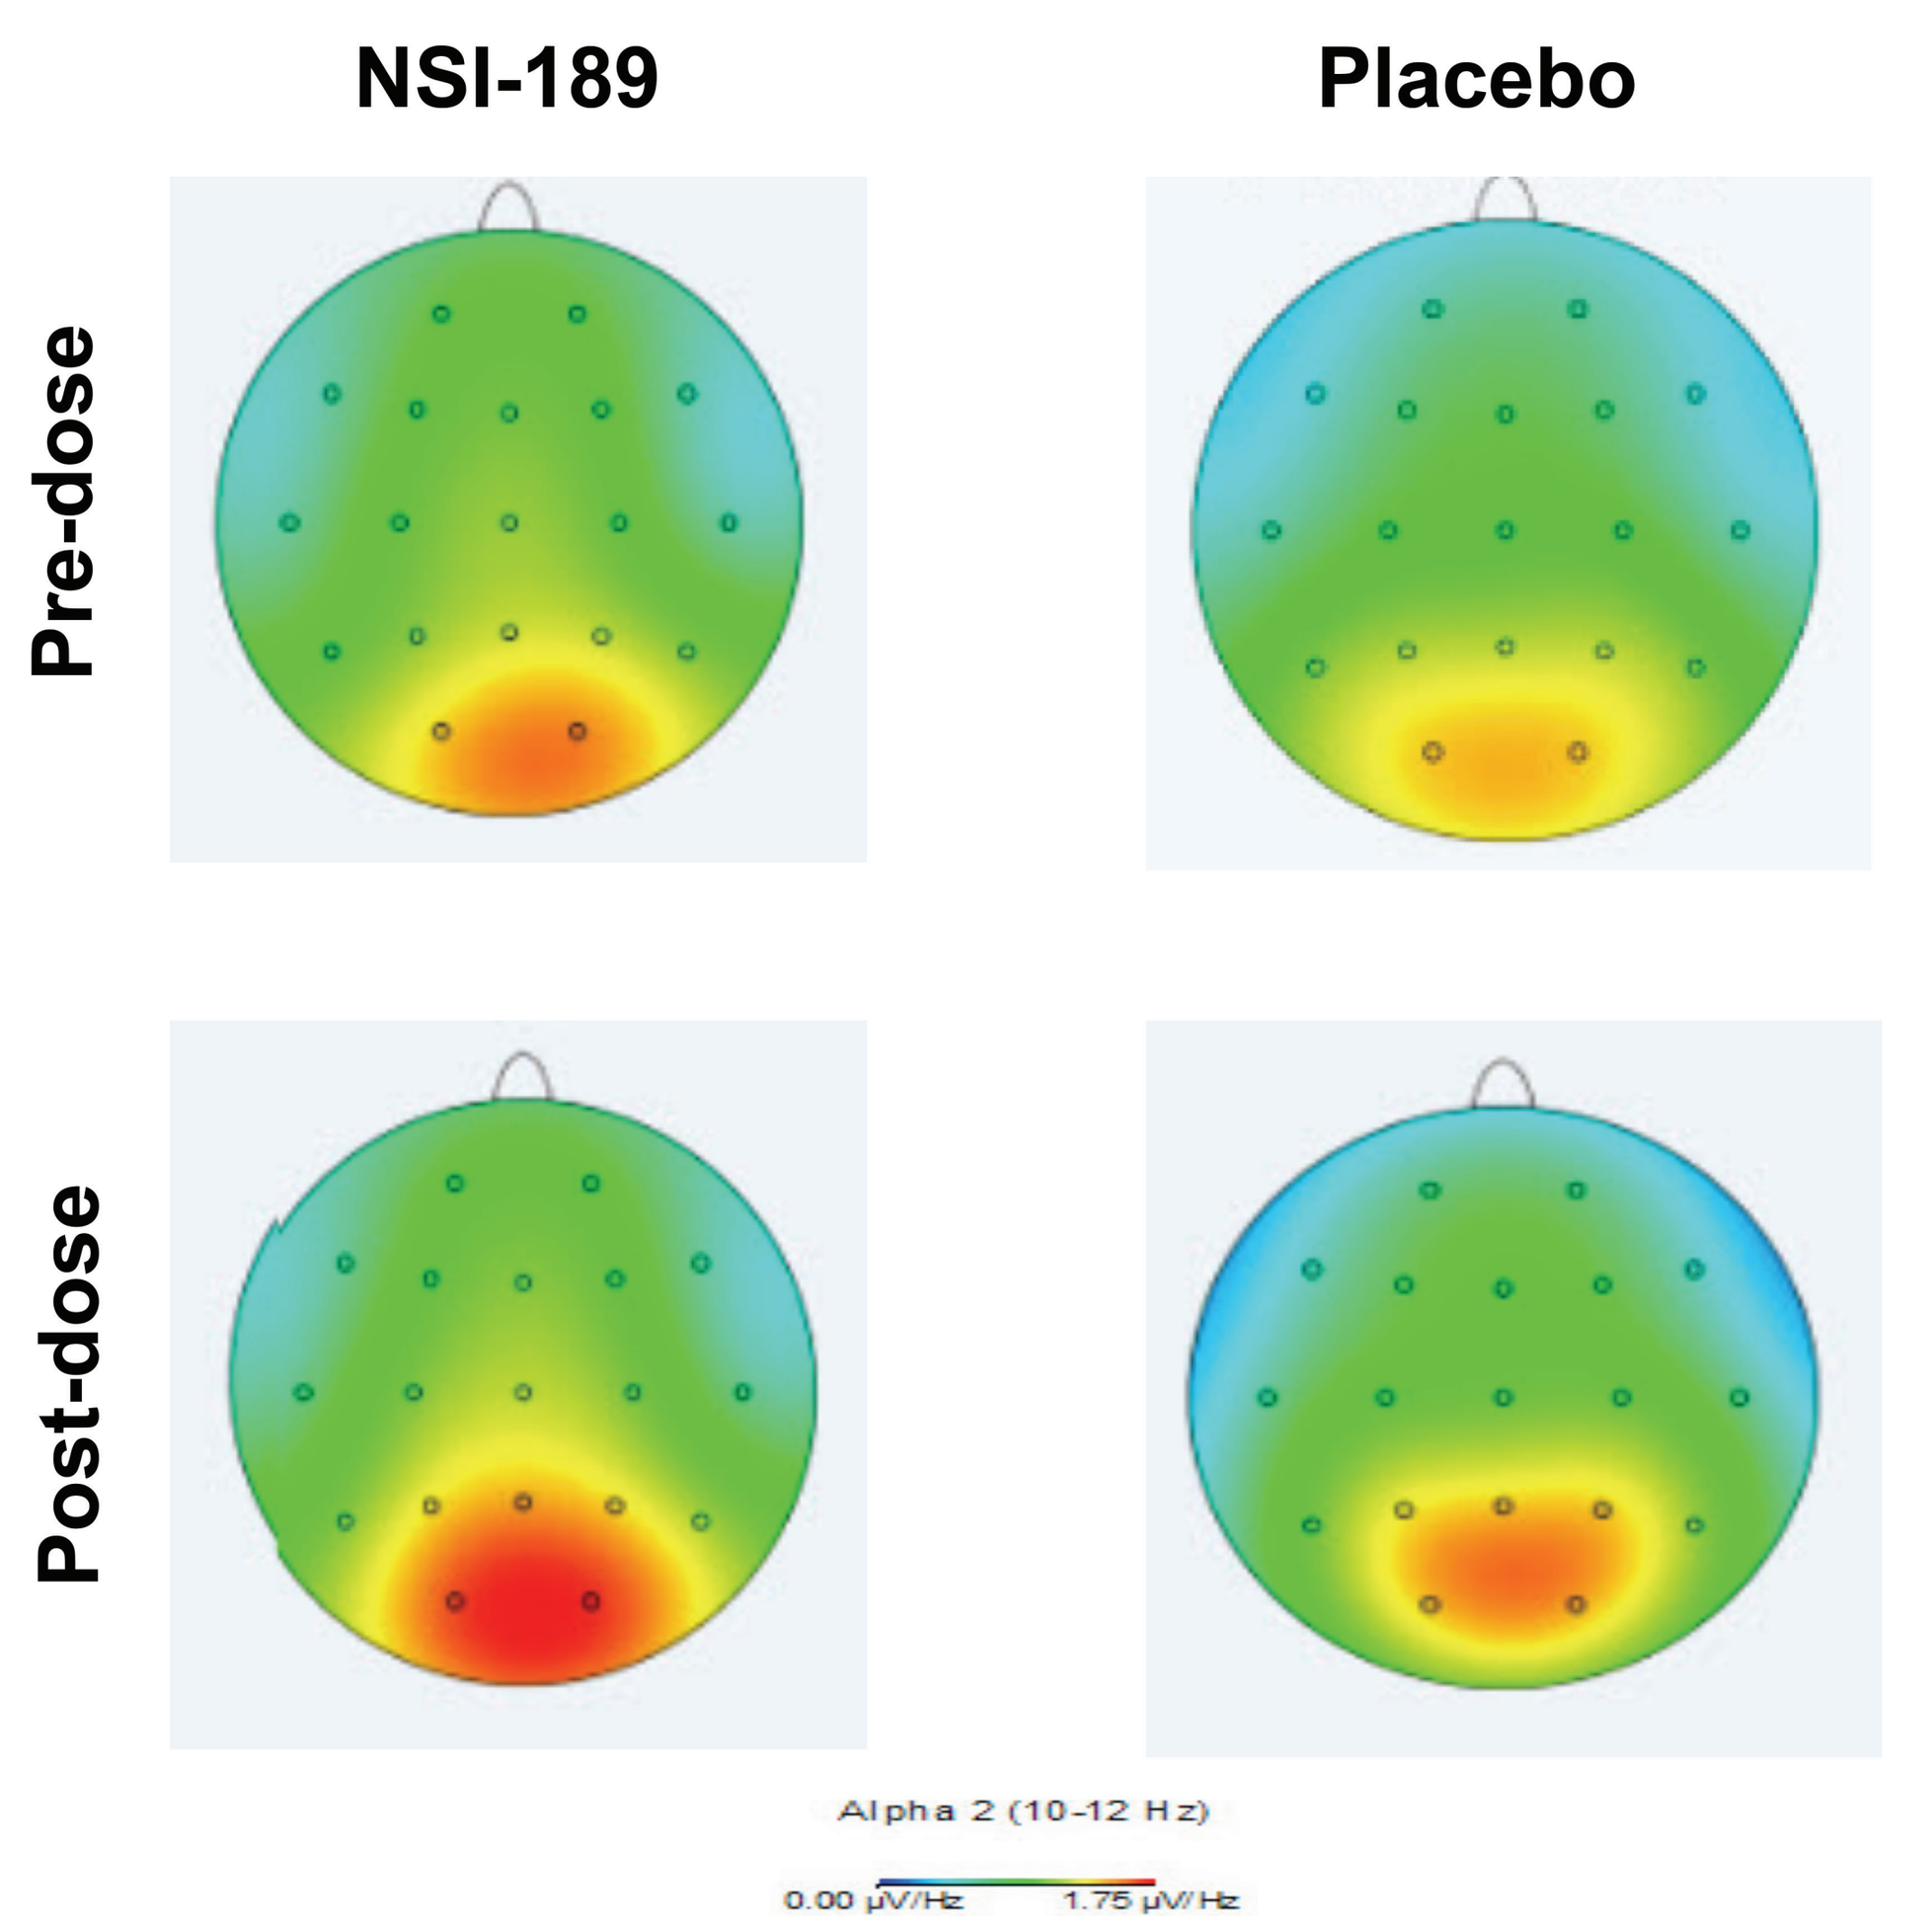
NSI-189
Placebo
Pre-dose
Post-dose
Alpha 2 (10-12 Hz)
0.00 µV/Hz
1.75 µV/Hz

Topographs of average amplitude at 10-12 Hz showing increased high-frequency (HF) alpha in patients receiving NSI-189 at Day 28. Differences scores comparing conditions show most significant differences between total subjects receiving NSI-189 (left) vs. Placebo (right) in the left posterior temporal and parietal regions.

Figure 3. Absolute difference in high-frequency alpha from Baseline at Day 14 and 28

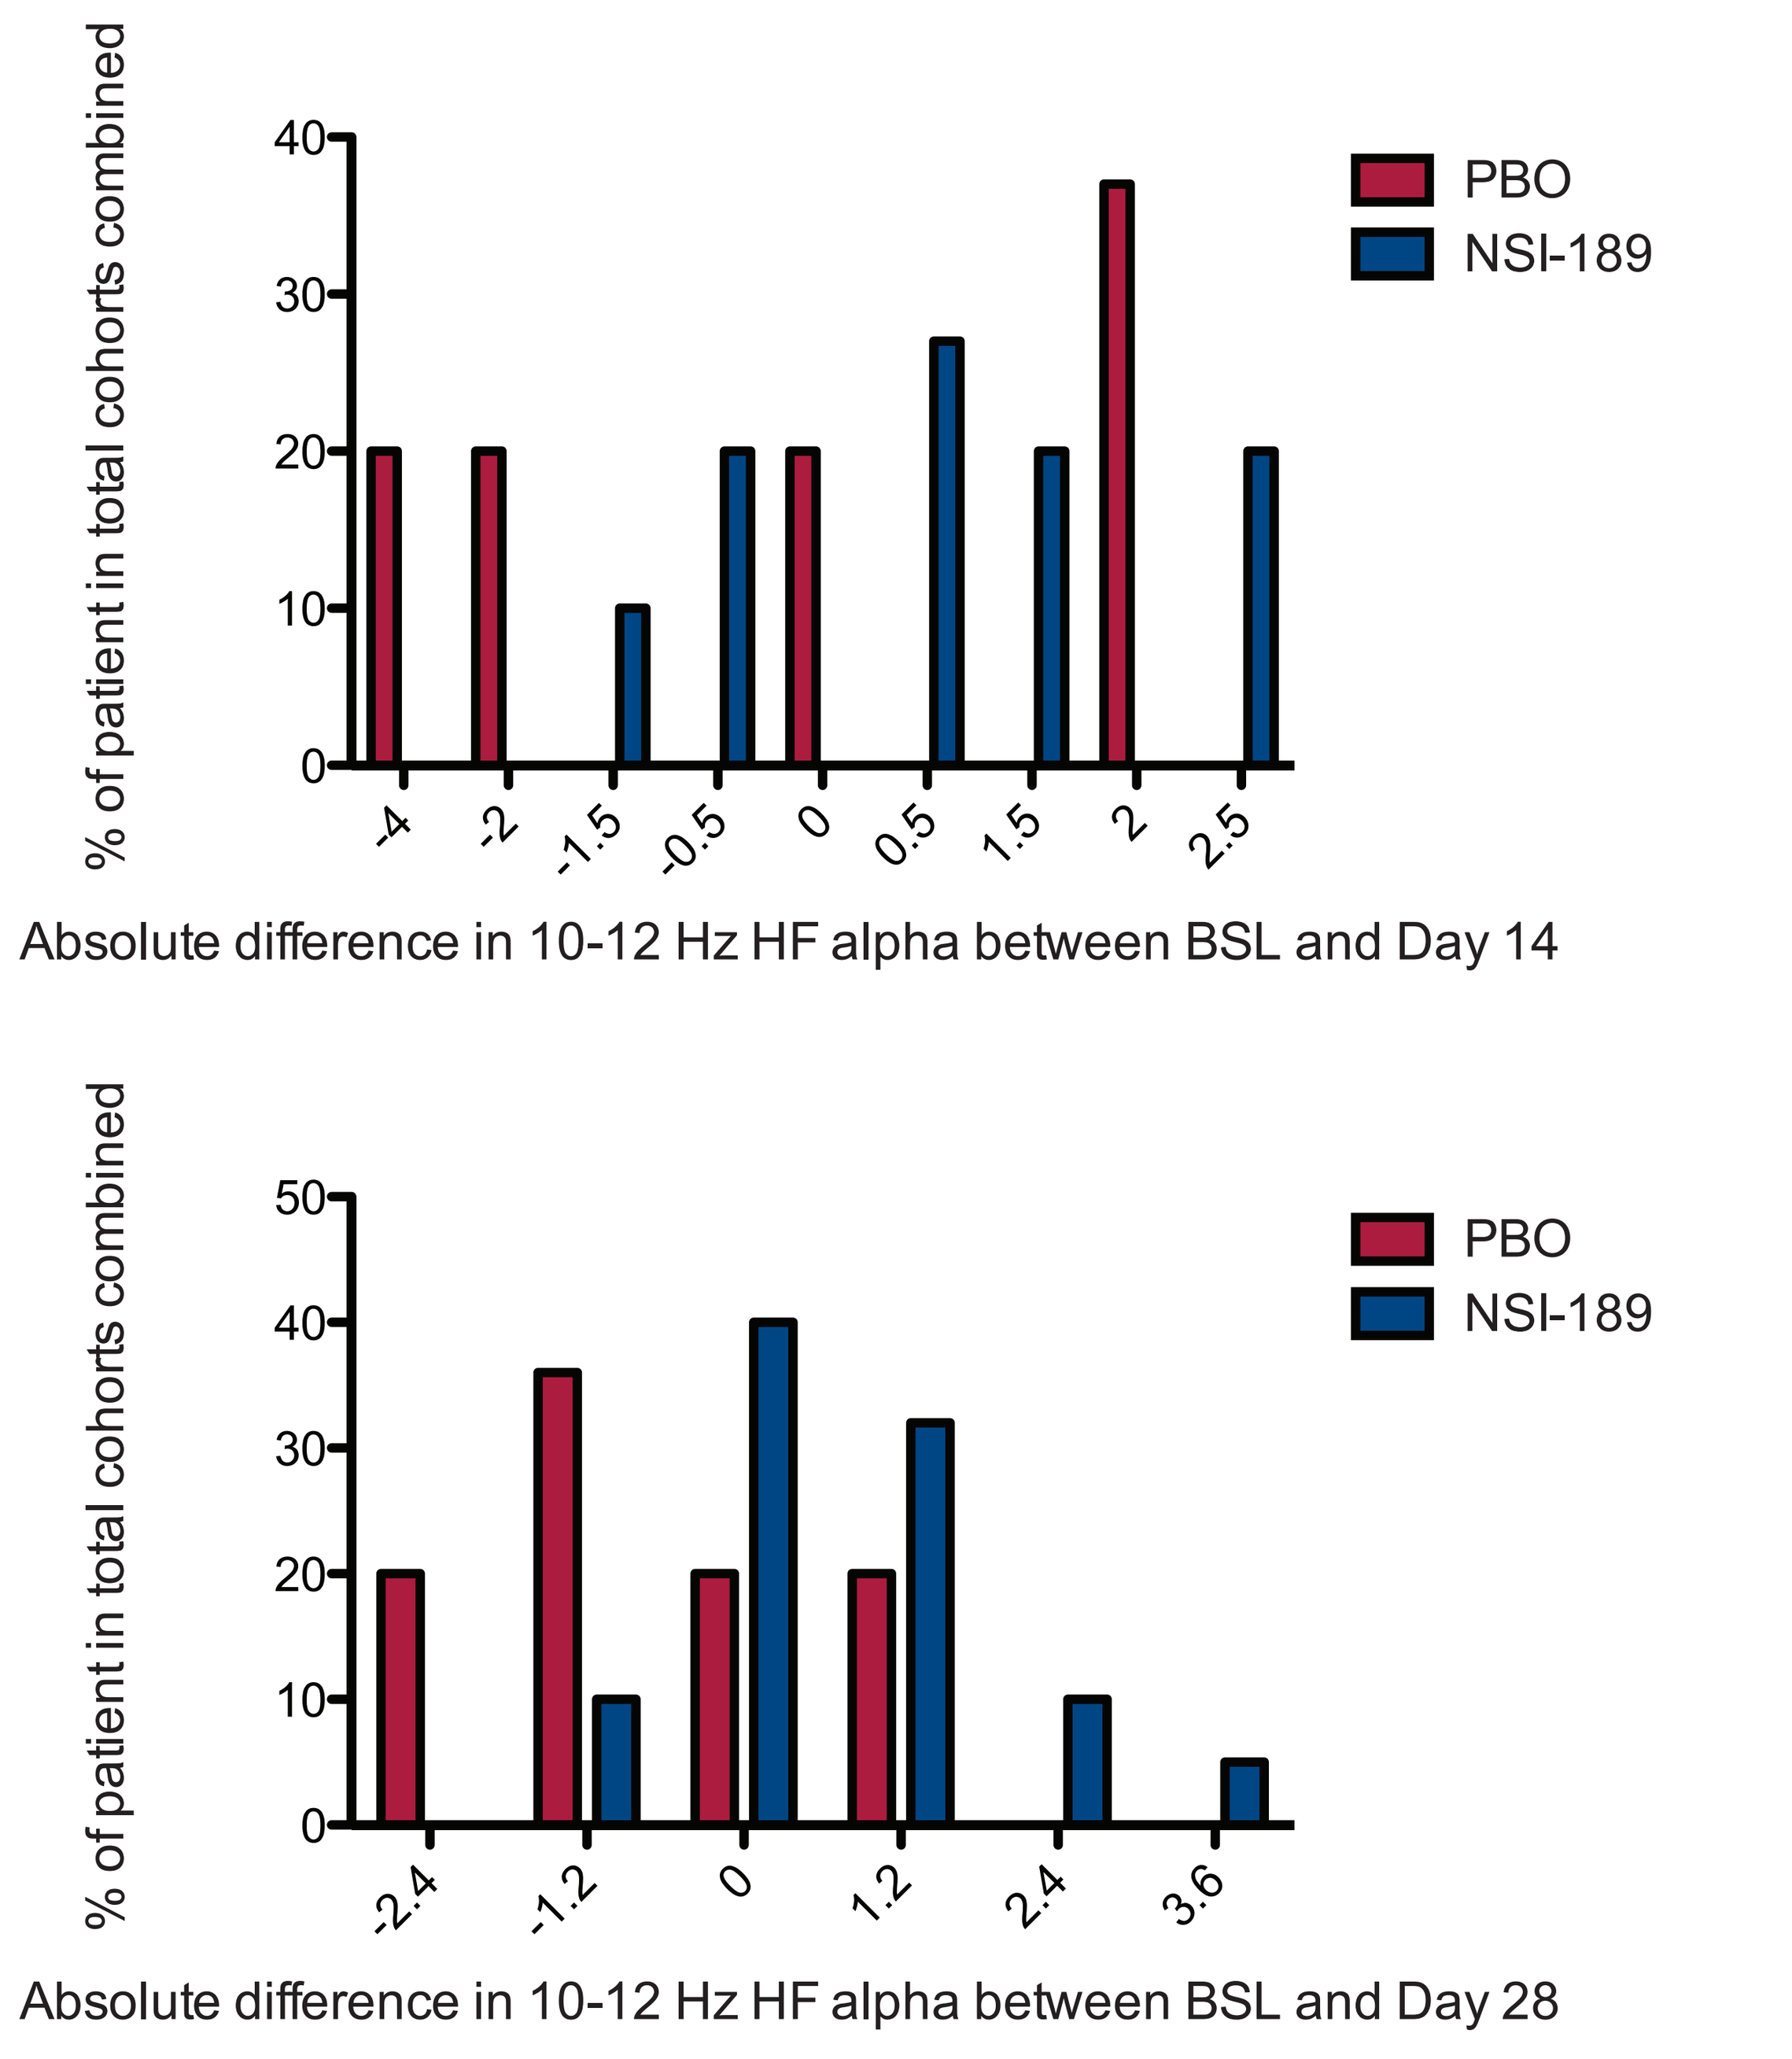
% of patient in total cohorts combined
40
30
20
10
0
-4
-2
-1.5
-0.5
0
0.5
1.5
2
2.5
PBO
NSI-189
Absolute difference in 10-12 Hz HF alpha between BSL and Day 14
% of patient in total cohorts combined
50
40
30
20
10
0
-2.4
-1.2
0
1.2
2.4
3.6
PBO
NSI-189
Absolute difference in 10-12 Hz HF alpha between BSL and Day 28

Percent of subjects (total cohorts) demonstrating absolute difference (Baseline – Day 14 or 28) in high-frequency (HF) alpha (10-12 Hz) recorded from the T5 (left posterior temporal lobe) electrode 6-hrs post-dose at Day 14 (top panel) and Day 28 (bottom panel) in patients receiving NSI-189. Treatment with NSI-189 demonstrates increases in HF alpha qEEG at Day 14 and 28. p < 0.03 (Day 14) and p < 0.02 (Day 28)

SUMMARY

1. There were no statistically significant demographic differences between patients with MDD randomized to receive either placebo, 40 mg QD, 40 mg BID, or 40 mg TID.
2. NSI-189 was generally well tolerated in patients with MDD.
3. Results of safety EEG assessments revealed no clinically significant EEG abnormalities associated with NSI-891 in patients with MDD across the 28-day dosing period.
4. qEEG analyses revealed increased high-frequency alpha frequency in the left posterior temporal (T5 electrode) and parietal (P3 electrode) regions in MDD patients receiving NSI-189 vs placebo.
5. Treatment with NSI-189 resulted in a higher proportion of patients with MDD demonstrating statistically significant increase in HF (10-12 Hz) alpha frequency at Day 14 and Day 28.
6. Analysis of coherence measures revealed increased prefrontal cortex coherence in MDD patients receiving NSI-189 vs placebo at Day 14 (data not shown).
7. Active treatment with NSI-189 produced decreased interhemispheric coherence compared to placebo (data not shown).

CONCLUSIONS

1. NSI-189, a novel compound with preclinical antidepressant properties was safely tolerated as monotherapy in patients with MDD.
2. NSI-189 produced qEEG changes in high-frequency alpha in the left posterior temporal and parietal regions in patients with MDD, consistent with data in depressed patients demonstrating treatment response to antidepressants with serotonergic activation (i.e., SSRIs).
3. NSI-189 may also exhibit pro-cognitive properties associated with increases in prefrontal alpha coherence.
4. Additional analyses will examine dose-response effects of NSI-189 vs placebo on qEEG parameters.

REFERENCES[1-8]

1. Hasin DS, Goodwin RD, Stinson FS et al., Epidemiology of major depressive disorder. *Arch Gen Psychiatry.* 2005;62:1097-1106.
2. Duman RS. Role of neurotrophic factors in the etiology and treament of mood disorders. *Neuromolecular Med.* 2004;5:11-25.
3. Boldrini M, Underwood MD, Hen R et al., Antidepressants increase neural progenitor cells in the human hippocampus. *Neuropsychopharmacol.* 2009;34:2376-89.
4. Tenke CE, Kayser J, Manna CG et al., Current source density measures of electroencephalographic alpha predict antidepressant treatment response. *Biol Psychiatry.* 2011;70:388-94.
5. Bruder GE, Sedoruk JP, Stewart JW et al., Electroencephalographic alpha measures predict therapeutic response to a selective serotonin reuptake inhibitor antidepressant: pre- and post-treatment findings. *Biol Psychiatry.* 2011;63:1171-77.
6. Tang SW, Helmeste D, Leonard B. Is neurogenesis relevant in depression and in the mechanism of antidepressant drug action? A critical review. *World J Biol Psychiatry.* 2012;13:402-12.
7. Wilson FJ, Leiser SC, Ivarsson M, et al., Can pharmaco-electroencephalography help improve survival of central nervous system drugs in early clinical development? *Drug Discov Today.* 2014;19:282-88.
8. Fava M, Johe K, Gertsik L et al., A phase 1b, randomized, double-blind, placebo-controlled, multiple-dose escalation study evaluating the effects of NSI-189 phosphate, a neurogenic compound, in patients with major depressive disorder (MDD) [abstract]. In: American Society of Clinical Psychopharmacology 2014 Annual Meeting; 2014 June 16-19; Hollywood, FL.: ASCP 2014, p.74.

CONTACT INFORMATION

Brett A. English, Pharm.D., Ph.D.
brett.english@PAREXEL.com

Larry Ereshefsky, Pharm.D., BCPP
larry.ereshefsky@PAREXEL.com

PAREXEL International
195 West Street
Waltham, MA 02451 USA
+ 1 781 487 9900
www.PAREXEL.com

Disclosures: BAE, MS and LE are full-time employees of PAREXEL International and receive financial compensation from numerous pharmaceutical companies for conduction of clinical trials. LG is a full-time employee of the California Clinical Trials Medical Group (CCTMG) and serves as Principal Investigator on industry-sponsored trials at PAREXEL. JJ is the CEO of Q-Metrx, Los Angeles and provides contract EEG/PSG services for PAREXEL. KJ is Chairman and Chief Scientific Officer of Neuralstem, Inc. MF, BH, MF, DM, GK and MF are full-time employees of the Clinical Trials Network and Institute (CTNI), Massachusetts General Hospital.