SYN-004 (ribaxamase) Protects the Gut Microbiome of Patients Treated with Ceftriaxone from Disruption and Reduces the Emergence of Antimicrobial Resistance

1337

John F. Kokai-Kun[1*], Charles Le[1] Ken Trout[1], Julia Cope[2], and Joseph Sliman[1]
[1]Synthetic Biologics, Inc., Rockville, MD, [2]Diversigen Inc. Houston, TX
jkokai-kun@syntheticbiologics.com

Synthetic BIOLOGICS

ABSTRACT

Background: When β-lactam antibiotics are administered intravenously, a significant portion of each dose can be excreted through the bile into the intestine. This excess antibiotic disrupts the balance of the gut microbiome making the recipient more susceptible to certain infections and can lead to the emergence of antimicrobial resistance. SYN-004 (ribaxamase) is an orally administered β-lactamase designed to be given with IV β-lactam antibiotics (penicillins and cephalosporins) to degrade excess antibiotics excreted into the upper GI tract before they can disrupt the gut microbiome and resistome.
Method: During a Phase 2b, clinical study with ribaxamase which demonstrated a significant reduction in Clostridium difficile infection in patients receiving ceftriaxone + ribaxamase, longitudinal fecal samples were collected from the patients. DNA extracted from these samples was 16S rRNA and whole genome sequenced, and the sequences were analyzed for changes in the gut microbiome and resistome. Statistical analyses were performed to determine correlations between changes in the gut microbiome and resistome and clinical study data.
Results: Sequence analyses revealed that ribaxamase protected the integrity of the gut microbiome, including preventing enterococcal mono-domination (defined as >30% of the microbiome being from one genus), and identified over 1300 AMR genes in the gut resistome. LefSe analysis of the gut resistome identified a family of β-lactamases (CfxA) and vancomycin resistance genes which demonstrated a significant increase in placebo-treated vs. ribaxamase-treated patients from pre-to post-antibiotics. Analysis by qPCR supported both new acquisition of these genes and expansion of existing AMR pools. Further statistical analyses demonstrated significant correlations between changes in the gut resistome and clinical study parameters including β-lactamase gene frequency and study drug assignment, and efflux pump gene frequency and vancomycin resistance.
Discussion: Taken together, these findings demonstrated that coadministration of ribaxamase with IV β-lactam antibiotics can protect the integrity of the gut microbiome and may help limit the emergence of AMR induced by these antibiotics.

BACKGROUND

- **The use of intravenous β-lactam antibiotics, including cephalosporins, are an important risk factor for the development of *Clostridium difficile* infection (CDI)**
- **SYN-004 (ribaxamase) is an orally administered β-lactamase (an enzyme of ~29kDa) designed to degrade excess IV β-lactam antibiotics excreted into the intestine**
- **Ribaxamase is intended to protect the gut microbiome from disruption and thus prevent CDI**
- **The use of ribaxamase may also have the added benefit of reducing the development of antibiotic resistance in the gut microbiota**

Ribaxamase degrades β-lactam antibiotics (including most penicillins and cephalosporins) by cleaving the β-lactam ring.

PHASE 2b POC STUDY

Patients admitted to the hospital for treatment of a lower respiratory tract infection

mITT = 412 patients

1:1

Ceftriaxone + Ribaxamase | **Ceftriaxone + Placebo**

+/- macrolides at the clinical site discretion

84 Multinational Clinical Sites

Phase 2b Clinical Study Schematic

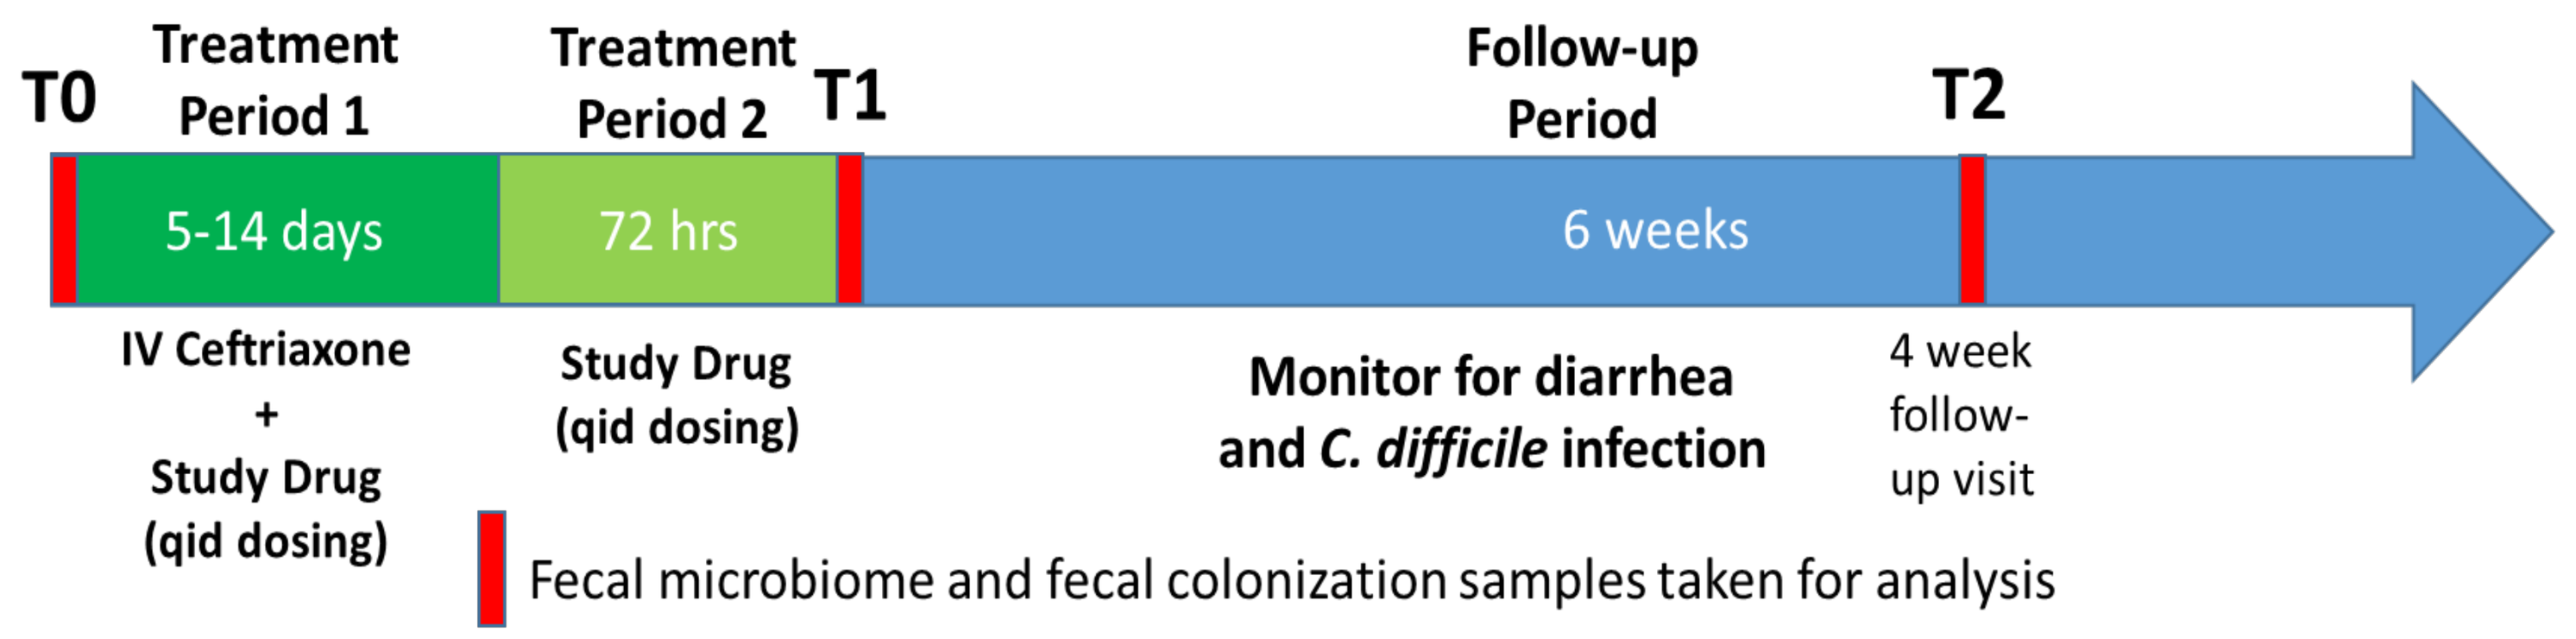
T0
Treatment Period 1
Treatment Period 2
T1
Follow-up Period
T2
5-14 days
72 hrs
6 weeks
IV Ceftriaxone + Study Drug (qid dosing)
Study Drug (qid dosing)
Monitor for diarrhea and C. difficile infection
4 week follow-up visit
Fecal microbiome and fecal colonization samples taken for analysis

Diarrhea = 3 or more loose or watery stools in a 24 hour period, samples are collected
CDI = local lab results for presence of *C. difficile* toxins A and/or B by an approved test (confirmed at a central lab by toxin ELISA)

Primary Endpoint:
Prevention of *C. difficile* infection (CDI)

Exploratory Endpoints:
Evaluate ability to limit disruption of the gut microbiome and prevent emergence of AMR

Phase 2b Clinical Study Results

- Ribaxamase **reduced the incidence of new onset CDI by 71%** as compared with placebo (7 vs. 2, confirmed at the central lab), p=0.045
- Ribaxamase appeared to be **well-tolerated** and **not affect the cure rate** for the primary infection
- Ribaxamase **reduced new colonization** with *C. difficile* and VRE and prevented enterococcal domination

Ribaxamase Protected the Gut Microbiome

- **652 Fecal Samples (229 patients) analyzed by 16S rRNA**
- **350 Fecal Samples (110 patients) analyzed by WGS**
- **100 Fecal Samples (38 patients) analyzed by qPCR**

Analysis of Changes in Beta Diversity

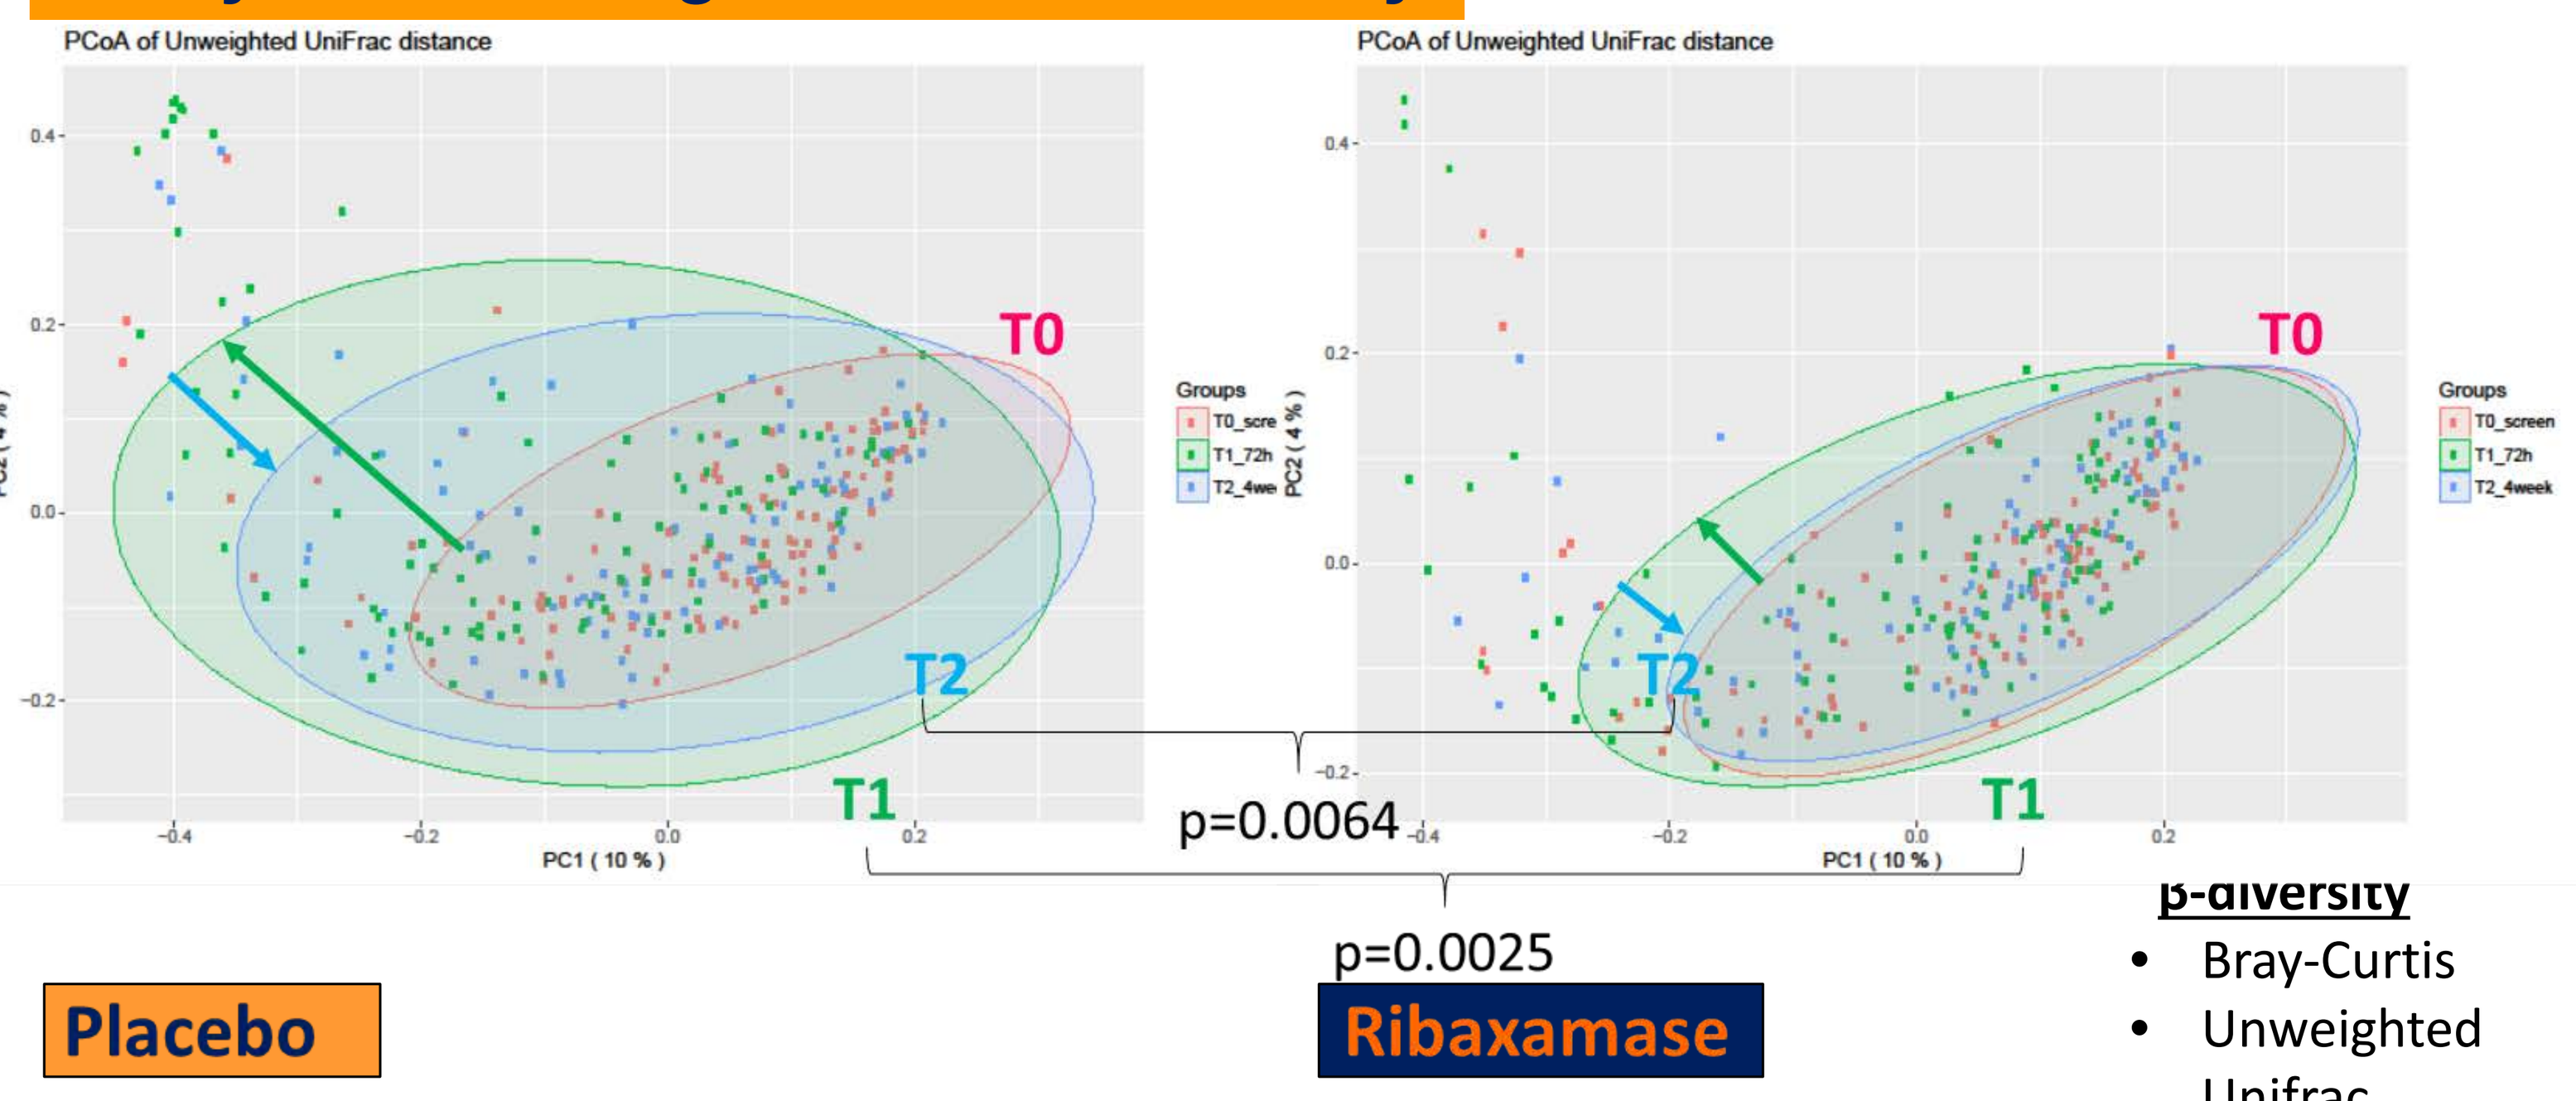
PCoA of Unweighted UniFrac distance
T0
T1
T2
Groups
T0_screen
T1_72h
T2_4week
PC1 (10 %)
PC2 (4 %)
p=0.0064
p=0.0025
Placebo
Ribaxamase

β-diversity
- Bray-Curtis
- Unweighted Unifrac

Beta diversity compares the community composition of two different sample sets

RESISTOME ANALYSIS-supported by the CDC

Analysis of the Change in Relative Abundance of AMR Genes

Collection point T0 to T1, Placebo vs. Ribaxamase-treated patients

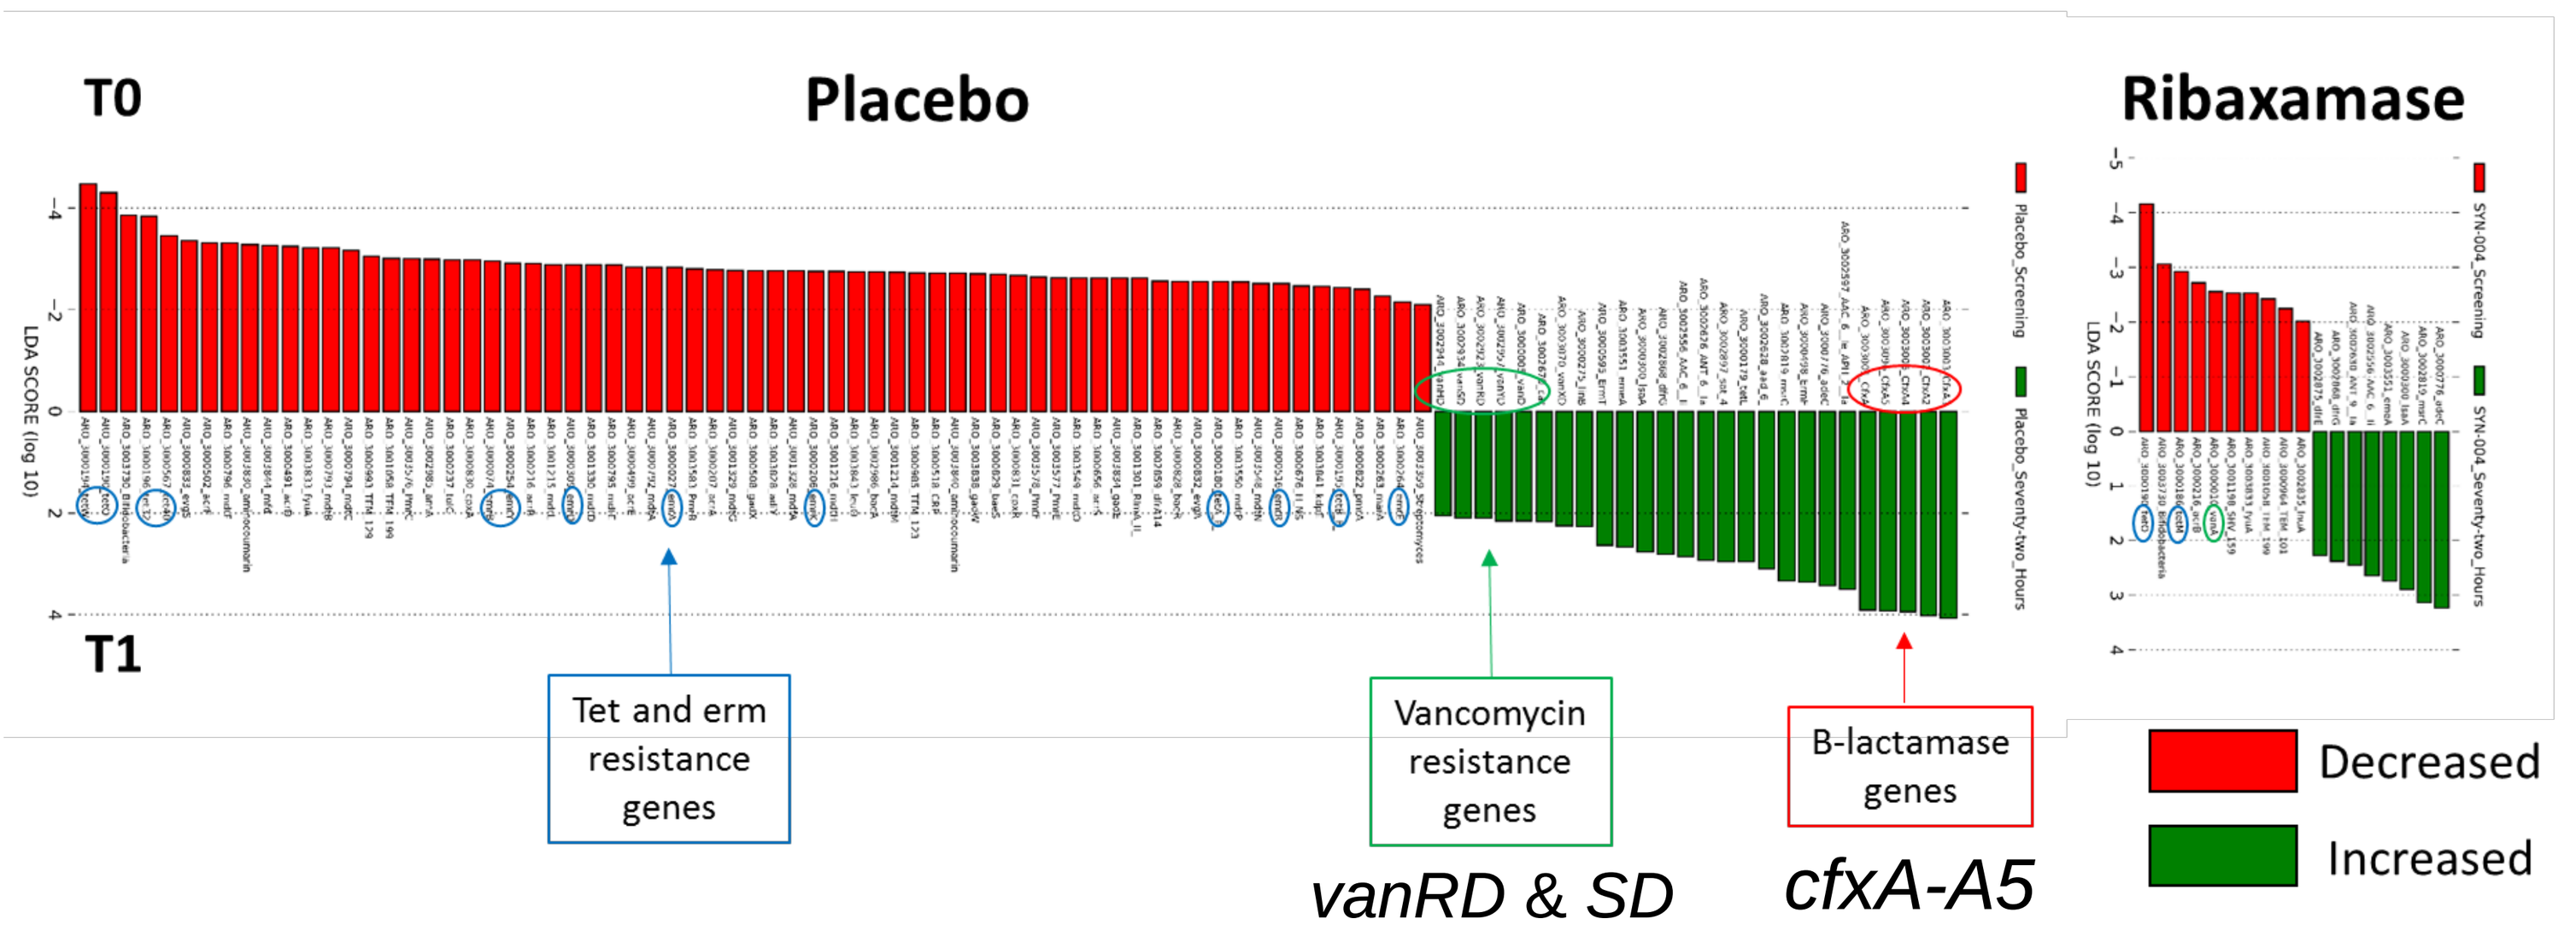
T0
T1
Placebo
Ribaxamase
LDA SCORE (log 10)
Tet and erm resistance genes
Vancomycin resistance genes
B-lactamase genes
vanRD & SD
cfxA-A5
Decreased
Increased

Identified significant increases in the CfxA family of β-lactamases and the VanSD/VanRD vancomycin resistance genes in placebo-treated group

- DNA extracted from **350 fecal samples** from the Phase 2b study sequenced by whole genome shotgun sequencing (Diversigen, Houston, TX)
- Interrogated against the **CARD database**
- 21,000,000 DNA matches
- **1300 AMR genes** identified with ~60,000 matches per sample
- Including many genes of interest, β-lactamases, vancomycin and macrolide resistance genes
- Statistical analysis (LefSE) was performed to determine which genes significantly changed from the screening sample (T0) to the post antibiotic sample (T1) in the placebo vs. the ribaxamase-treated patients

qPCR Analysis of *cfxA* and *vanRD*

Comparison of Copy Number vs. Relative Abundance

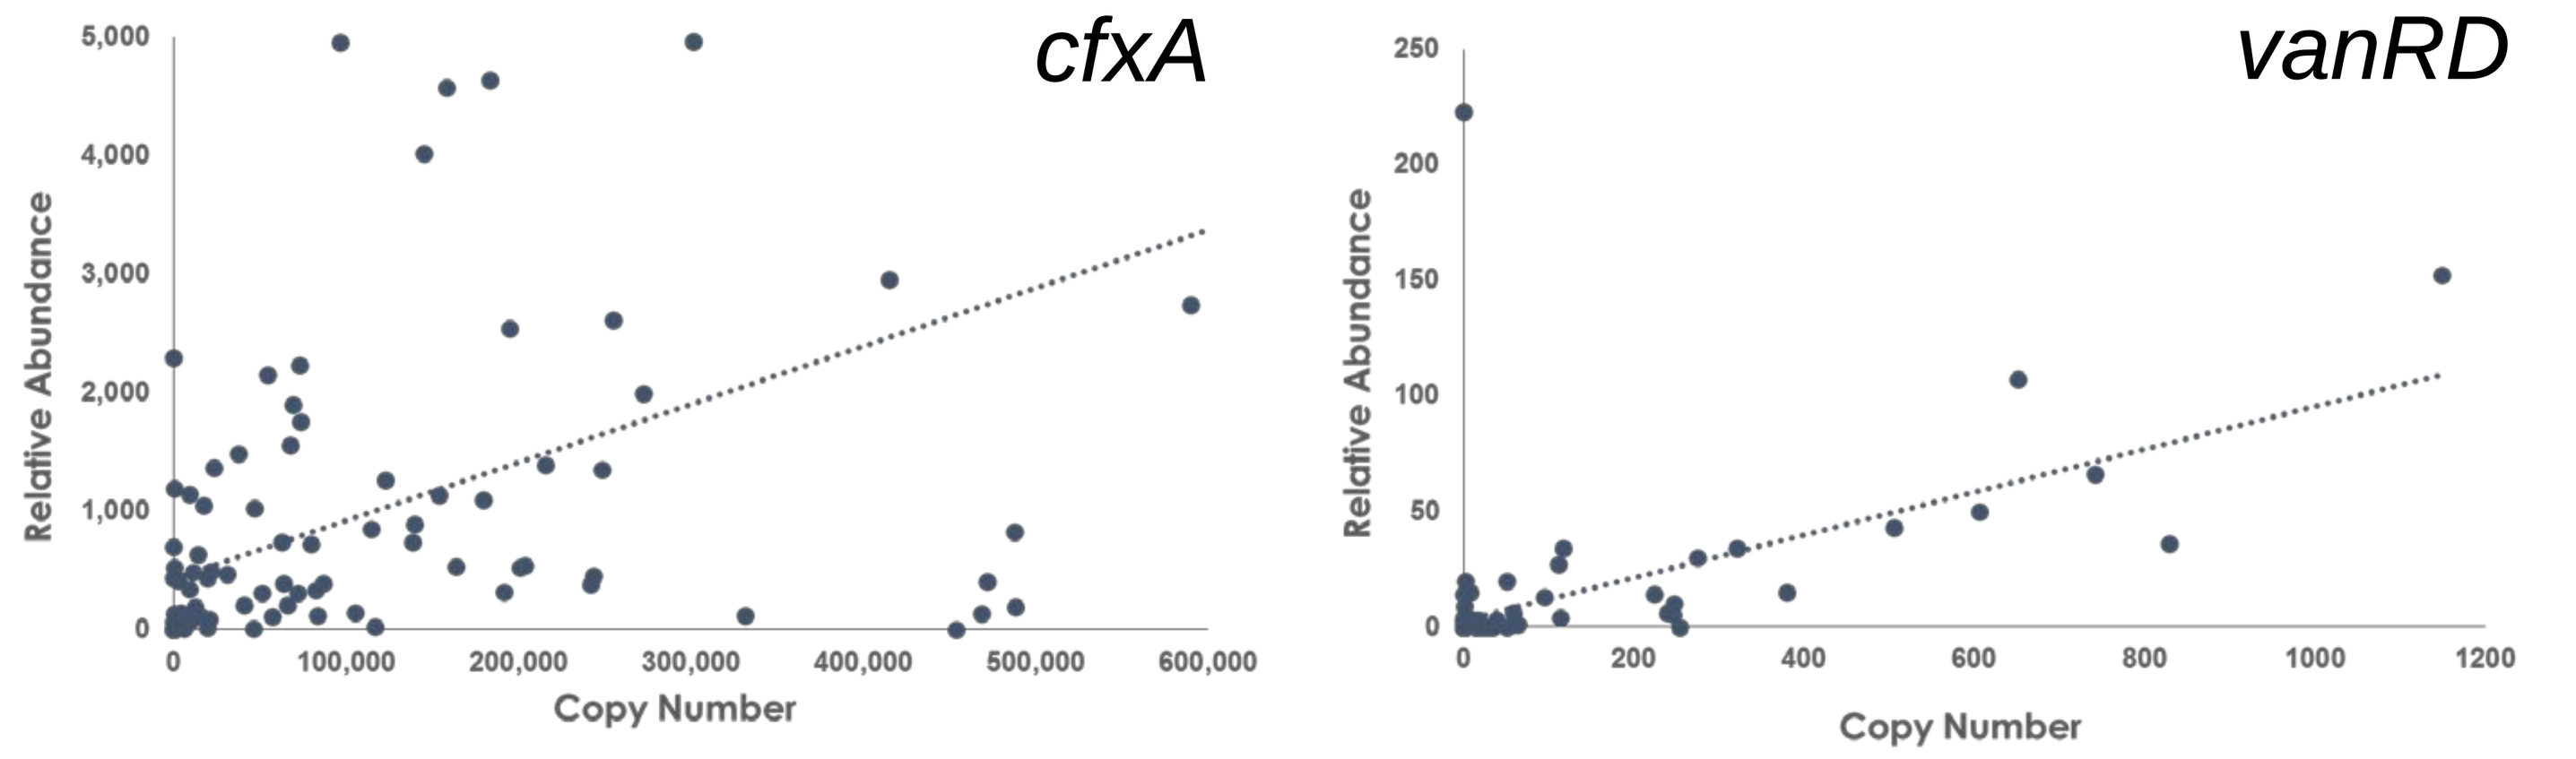
cfxA
vanRD
Relative Abundance
Copy Number

Relative abundance and copy number appeared to demonstrate fairly good concordance

qPCR results were consistent with both new acquisition of resistance and expansion of existing resistance

Supported by: CDC contract 200-2016-91935

Change in Copy Number from the T0 Screening Sample

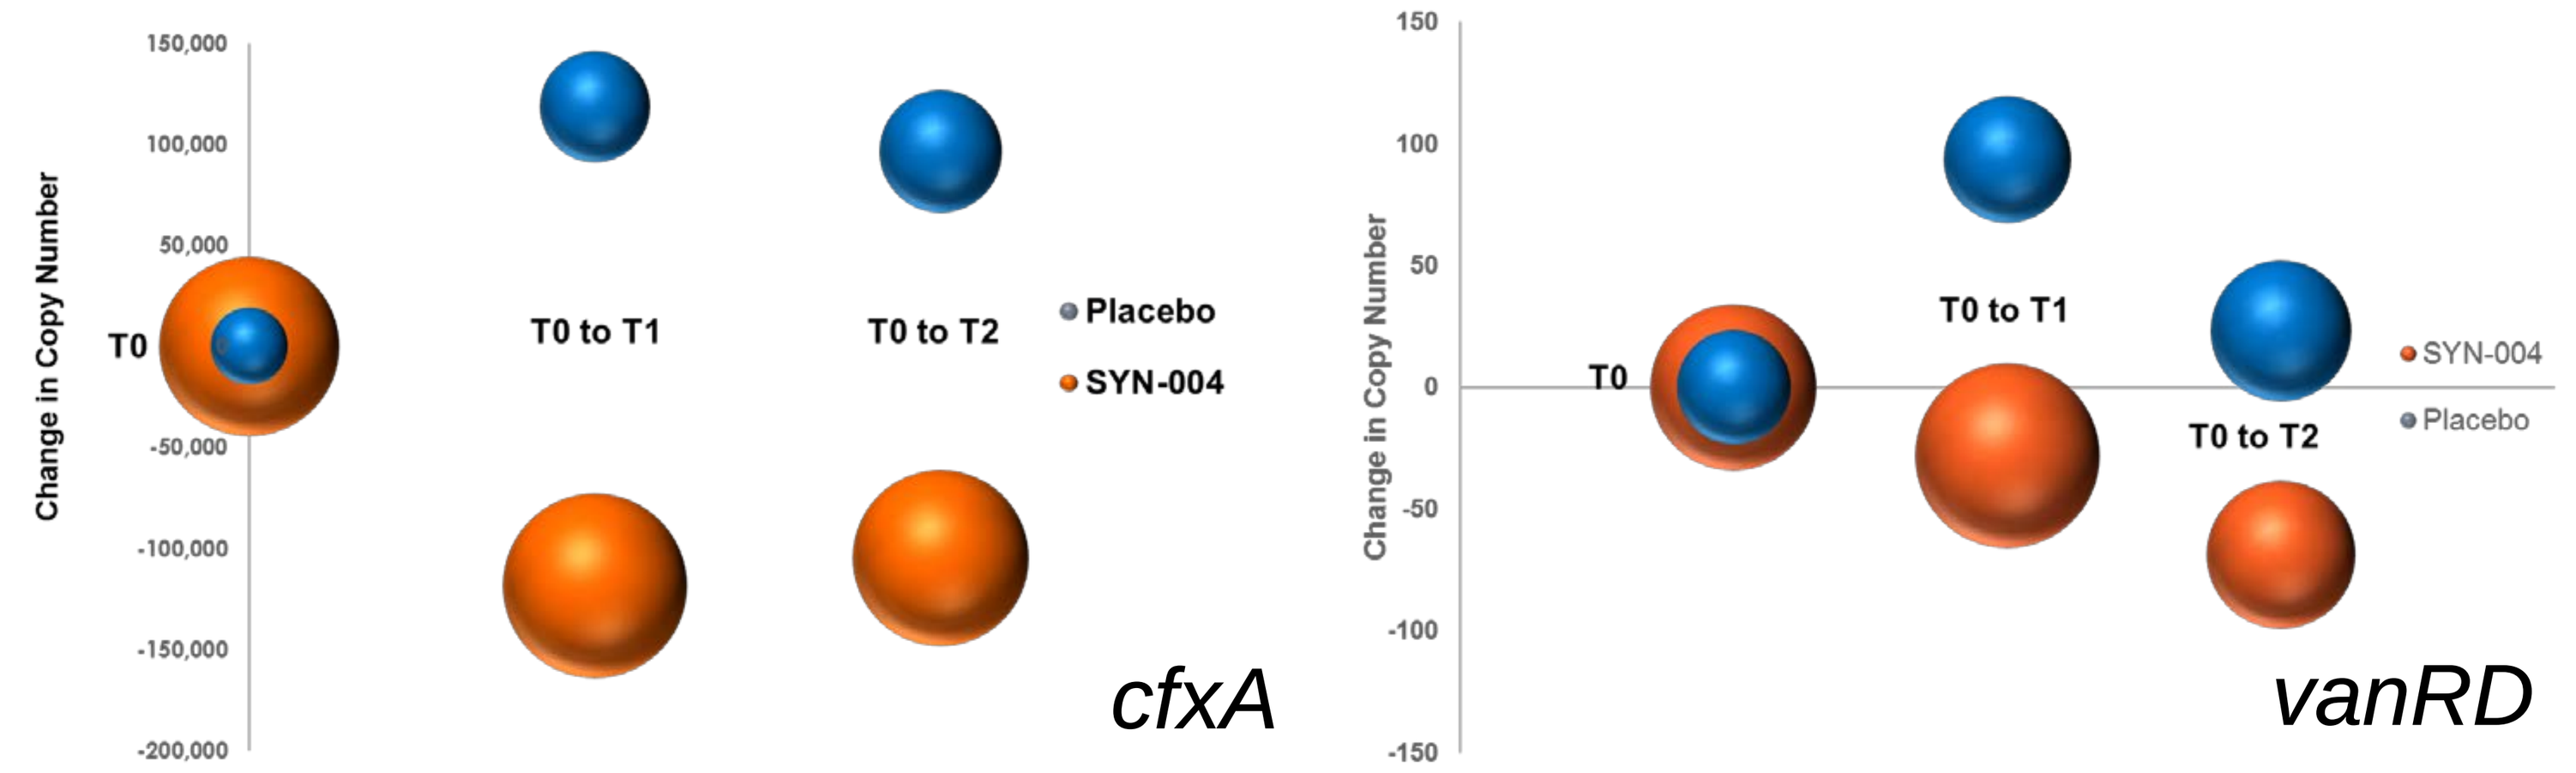
Change in Copy Number
T0
T0 to T1
T0 to T2
Placebo
SYN-004
cfxA
vanRD

Mean change in copy number from screening with the value for the screening samples set to zero. The copy number for *cfxA* or *vanRD* at the T1 and T2 collection points as determined by qPCR were subtracted from the copy number at T0 for each patient. The mean change was then calculated. The size of each ball represents the standard error for each sample set.

Correlation of Antibiotic Resistance Data with Clinical Data

Parameter (number of AMR genes ID'd)	β-lactamases (781)	Vancomycin (34)	Macrolide (8)	Efflux Pumps (39)
Study Drug (SYN-004/placebo)	PBO 8* + [1]	PBO 11 + [6]		SYN 36 +
New Colonization with VRE	nd**	22 + [7]	nd	nd
Enterococcal Monodomination	nd	25 + [8]	nd	nd
Colonization with ESBL-GNB	55 + [2]	nd	nd	nd
Use of Proton Pump Inhibitors	6 + [3] 3 − [4]			
Use of Macrolides	2 +		1 +	35 + [9]
No Food by Mouth (NPO)		7 + [9]		32 + [9]
Study Endpoint of CDI or AAD	44 +			2 + [10]
Increasing Age	5 + [5] 2 −			
Gender (Male [M]/Female [F])	F1 +	M6 + [9] F1 +		

\+ Positive correlation − Negative correlation *Number of genes
**Not determined

1-Includes the *cfxA-A5* genes
2-Includes TEM, OXA, OXY, ACT and CTX
3-Includes the *cfxA-A5* genes
4-Includes *cfxA6*
5-*cepA* only widely distributed gene
6-Includes *vanA* and *vanD* (*vanRD* & *SD*)
7-Includes *van A, B, C* & *G*
8-Includes *van A, B, D* & *G*
9-Includes the *vanB* operon
9-Overlaps the 36 for study drug
10-These 2 genes were 2 of 4 that did not overlap between Study Drug and NPO

A linear model was used to compare the **change in the relative abundance** of each AMR gene in each subclass from the T0 to the T1 collection point. Analyses were conducted based on the log transformed values. For each gene, both the t-test and Wilcoxon test were used for the comparisons between the groups, and nominal P-values were provided from these tests without multiplicity adjustment. A linear regression was used to examine the impacts of potential factors on the change from baseline (T0) at 72 hours (T1) for each gene in each subset. The correlation between colonization with certain AMR species and the presence of specific AMR genes was determined using a *t*-test. For each gene, the *t*-test compared the means of log transformed values between the two groups

CONCLUSION

Ribaxamase reduced ceftriaxone-mediated changes in the gut microbiome and gut resistome and has the potential to reduce the emergence of AMR

Synthetic Biologics, Inc.