GAL-101 prevents β-amyloid induced membrane depolarization in two different types of retinal cells

#560.05 / C90

Parsons, C.G.[1], Mazzanti, M.[2], Gornati, S.V.[3], Pizzi, E.[4] and Rammes, G.[5]

[1]Galimedix, Kensington, MD; [2]Univ. di Milano, Milano, Italy; [3]Erasmus MC, Rotterdam, Netherlands; [4]Univ. of Milan, Milano, Italy; [5]Clin. Rechts der Isar, Munich, Germany

Soc Neurosci Abstr. 49th Society for Neuroscience Annual Meeting. Chicago, USA, 19-23rd October 2019.

Abstract

Glaucoma and age-related macular degeneration (AMD) are two of the major causes of progressive loss of vision and ultimate blindness worldwide. Retinopathies share several pathological features with Alzheimer´s disease (AD). Among other neurodegenerative events, in AD abnormalities in the retina such as visual function impairment, optic neuropathy, retinal ganglion cell (RGC) loss, decreased number of optic nerve fibres, astrocytosis and activation of immune competent microglia and Muller cells are evident. AD as well as glaucoma and AMD are characterized by the presence of elevated concentrations of amyloid beta (Aβ) peptide. Under such conditions $Aβ_{1-42}$ has the tendency to aggregate forming toxic soluble oligomers that are now considered to be the most harmful amyloid species. One strategy adopted to prevent cell damage caused by these soluble toxic oligomers is to impair their aggregation.

In the present work, using electrophysiological patch clamp measurements, we show that exposure of acutely isolated retinal cells to $Aβ_{1-42}$ caused a strong depolarization of the resting potential of the plasma membrane. The average resting membrane potential of cultured RGCs and retinal pigment epithelium (RPE) cells was -68.3 ± 1.9 mV and -40.2 ± 2.4 mV respectively. Acute application of $Aβ_{1-42}$ depolarized the membrane potential of RGCs with an EC_{50} of 44.1 ± 6.3 nM and a maximal effect at 100 nM to around -10 mV. Higher concentrations of $Aβ_{1-42}$ were required to cause depolarization of RPE cells (EC_{50} = 790 ± 80 nM).

GAL-101 is a small D-amino acid dipeptide that modulates $Aβ_{1-42}$ aggregation by triggering a non-amyloidogenic aggregation pathway and thereby reduces the amount of intermediate toxic soluble oligomeric $Aβ_{1-42}$ species. Pre-incubation of RGCs for one hour with a 1:20 ratio was able to stabilize the cell resting potential to around -50 mV. Similar effects were seen with a 1:20 ratio of GAL-101 to $Aβ_{1-42}$ for RPE cells.

In the therapeutic situation one could expect more subtle changes over more prolonged periods of time due to much lower chronically elevated concentrations of $Aβ_{1-42}$. Stoichiometric requirements for GAL-101 would therefore rather be superseded by its true affinity for $Aβ_{1-42}$ of around 30 nM.

Aβ: a New & Important AMD target

Author et al.	Year	Journal	Vol	Pages	Highlight - First Citation
Bradt, B.M.	1998	J Exp Med	188	431-8	Aβ causes key phenotypes of clinical AMD: activation of complement
Mullins, R.F.	2000	FASEB J	14	835-46	In human eyes Aβ is detected in drusen
Dentchev, T.	2003	Mol Vis	9	184-90	In human eyes Aβ-staining is co-localized with geographic atrophy (GA) atrophy
Yoshida, T.	2005	J Clin Invest	115	2793-800	Aβ causes key phenotypes of clinical AMD: activation of VEGF
Malek, G.	2005	Proc Natl Acad Sci.	102	11900-5	Senescent C57BL/6 mice fed a high cholesterol diet developed sub-RPE Aβ deposits
Luibl, V.	2006	J Clin Invest	116	378-85	In human eyes **oligomeric** Aβ is detected in drusen
Ding, J.D.	2008	Vision Res	48	339-45	In mouse models of AMD systemic anti-Aβ antibodies protect from visual deficits
Bruban, J.	2009	Aging Cell	8	162-77	Aβ is toxic to RPE cells *in vitro*
Tsuruma, K.	2010	J Neurochem	113	1545-54	Aß is toxic to RGCs *in vitro*
Ratnayaka, J.A.	2015	Eye (Lond)	29	1013-26	Subretinal injection of nanomolar oligomeric Aβ in mice produces an AMD phenotype
Guymer, R.	2015	Arch Clin Exp Ophthalmol	253	1347-54	Aβ recognized as biomarker in diagnosing AMD
Zhao, T.	2015	J Neuroinflammation	12	121	In animal models of AMD, retinal Aβ accumulates with the same distribution as in AMD patients
Lynn, S.A.	2017	Neural Regen Res	12	538-548	Extensive review on the role of Aß in AMD - focus on subretinal injection of nanomolar oligomeric Aβ

Aβ: a New & Important Glaucoma target

Author et al.	Year	Journal	Vol	Pages	Highlight - First Citation
McKinnon, S.J.	2002	Invest Ophthalmol Vis Sci	43	1077-87	In rat models of glaucoma Aβ is detected in RGCs and vicinity and is associated with extensive RGC death
Walsh, D.T.	2005	Neuropathol Appl Neurobiol	31	491-502	Intravitreal injection of Aβ causes RGC apoptosis and cell death
Goldblum, D.	2007	Invest Ophthalmol Vis Sci	48	5085-90	Aβ in RGCs and optic nerve is increased with age in a spontaneous, chronic age-related mouse model of glaucoma
Guo, L.	2007	Proc Natl Acad Sci U S A	104	13444-9	Apoptosis in the Morrison rat glaucoma model was significantly reduced by an Aß antibody and an Aß aggregation inhibitor
Bruban, J.	2009	Aging Cell	8	162-77	Aß is toxic to RPEs *in vitro*
Kipfer-Kauer, A.	2010	Curr Eye Res	35	828-34	In mouse models of glaucoma Aβ is detected in RGCs and vicinity and is associated with extensive RGC death
Tsuruma, K.	2010	J Neurochem	113	1545-54	Aß is toxic to RGCs *in vitro*
Böhm, N.	2012	Poster			Aβ is significantly increased in RGCs of glaucoma patients
Salt, T.E.	2014	Neurotox Res	26	440-6	In the Morrison rat glaucoma model MRZ-99030 reduces apoptosis by 60% and cell death by 95%
Ratnayaka, J.A.	2015	Eye (Lond)	29	1013-26	Subretinal injection of nanomolar oligomeric Aβ in mice produces an AMD phenotype
Mohd Lazaldin, M.A.	2018	Int J Neurosci		1-14	Intravitreal injection of Aβ causes RGC apoptosis, pathological changes in morphology and cell death in a time and dose-dependent manner

Resting membrane potential measurements in Retinal Ganglion Cells (RGCs).

Resting membrane potential measurements in Retinal Ganglion Cells (RGCs)

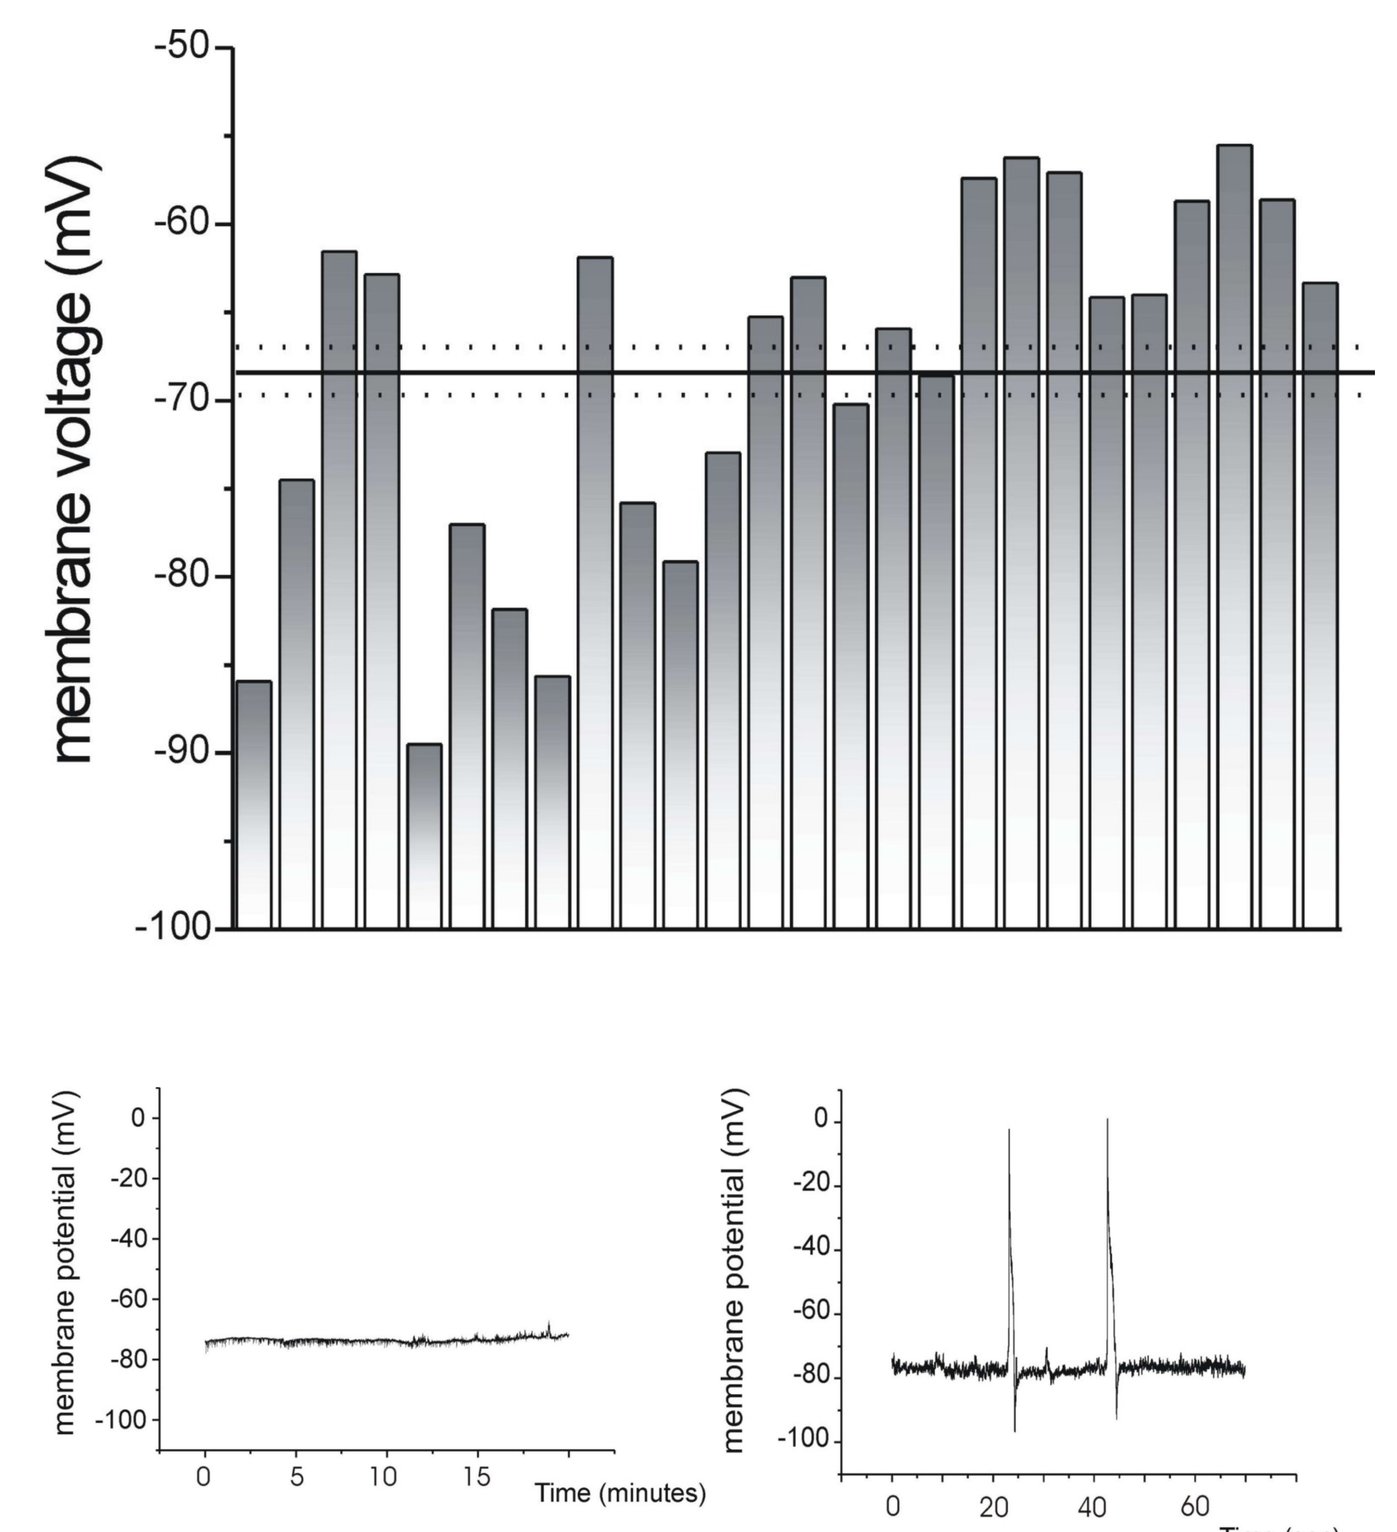
membrane voltage (mV)
membrane potential (mV)
Time (minutes)
Time (sec)

RGCs membrane potential concentration / response during $Aβ_{1-42}$ incubation

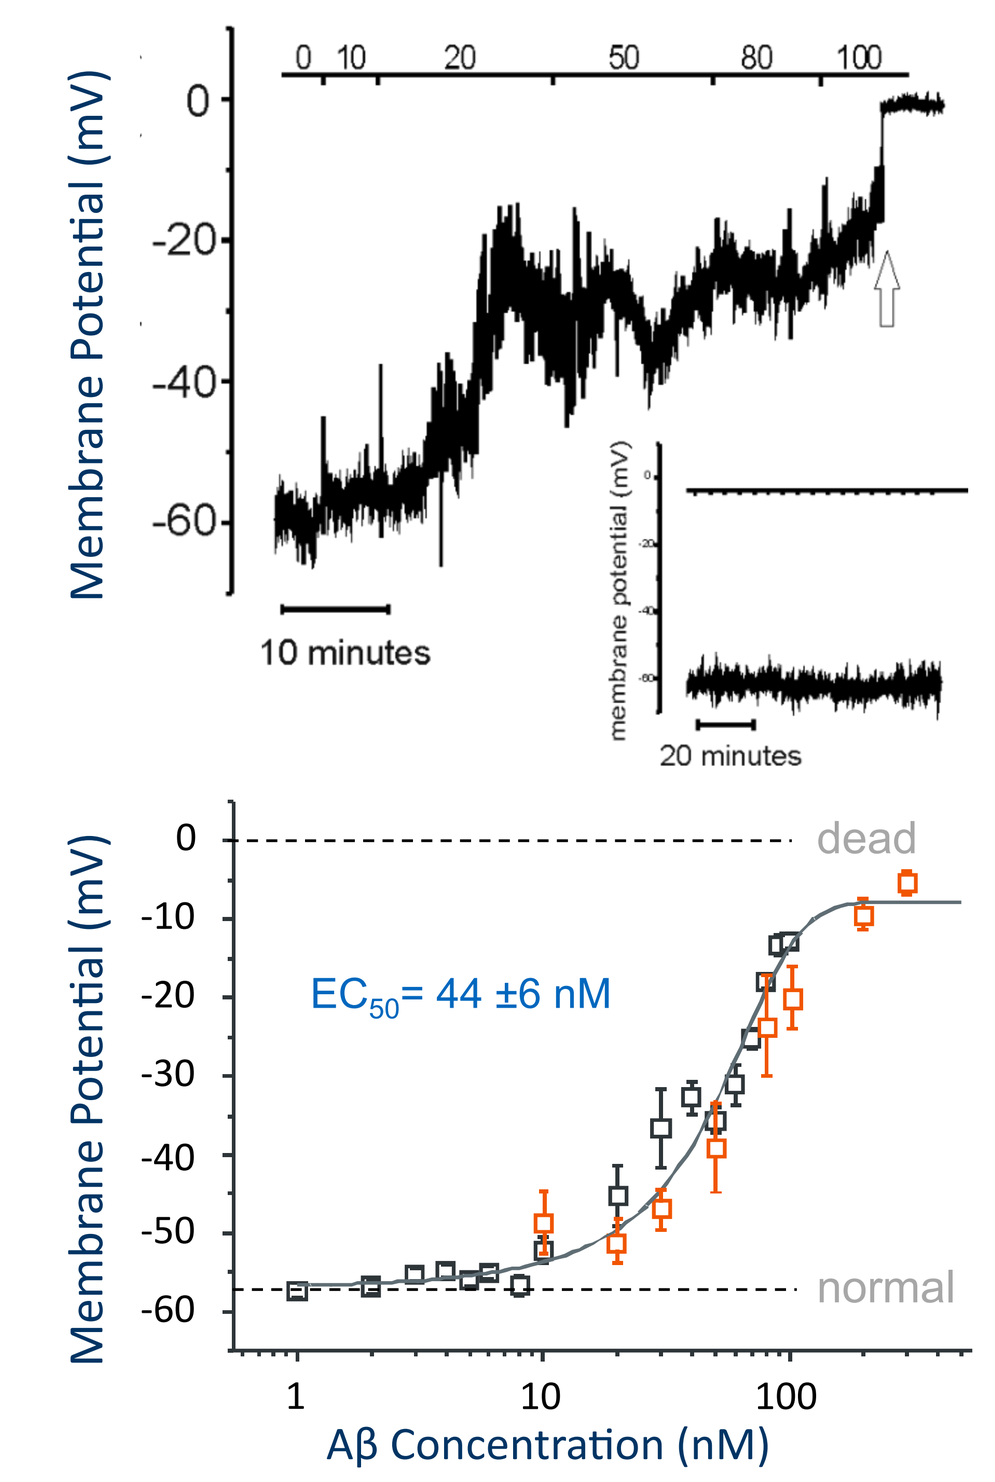
Membrane Potential (mV)
10 minutes
20 minutes
dead
normal
EC_{50}= 44 ±6 nM
Aβ Concentration (nM)

GAL-101 prevents RGCs membrane depolarization induced by $Aβ_{1-42}$

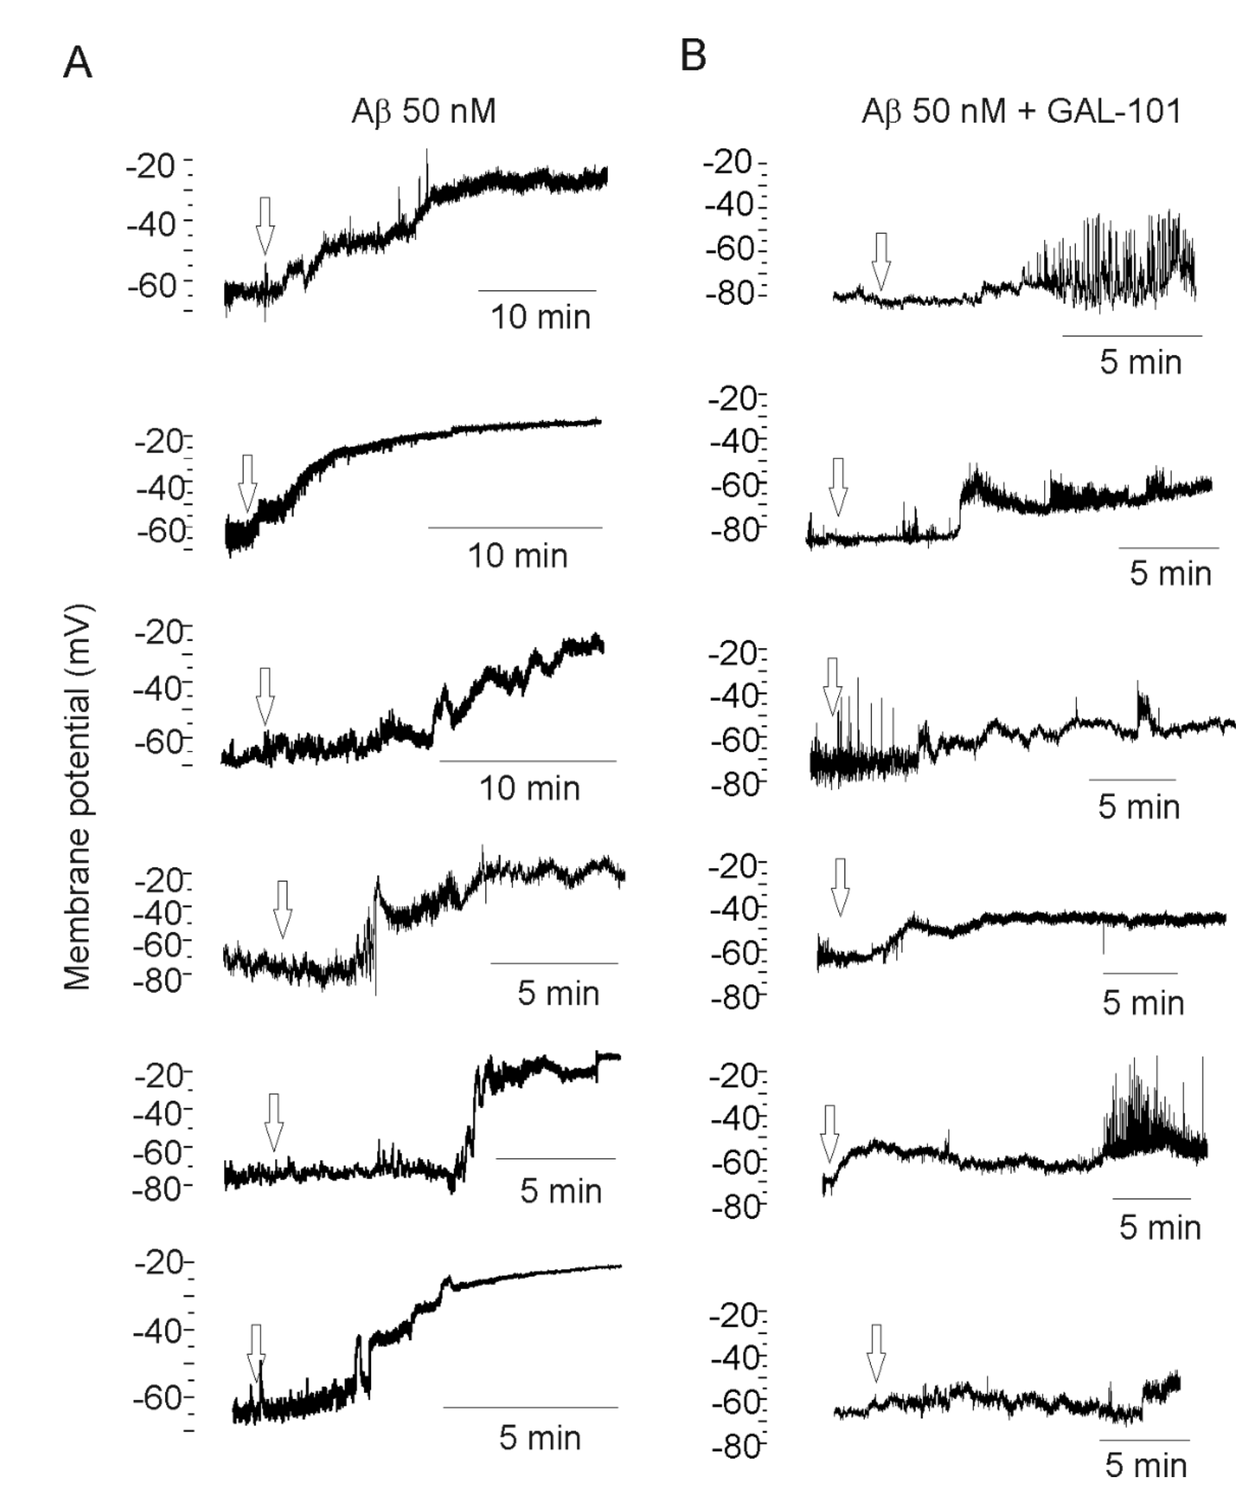
A
B
Aβ 50 nM
Aβ 50 nM + GAL-101
Membrane potential (mV)
10 min
5 min

Quantification of GAL-101 action on RGCs membrane depolarization induced by $Aβ_{1-42}$ 50 nM

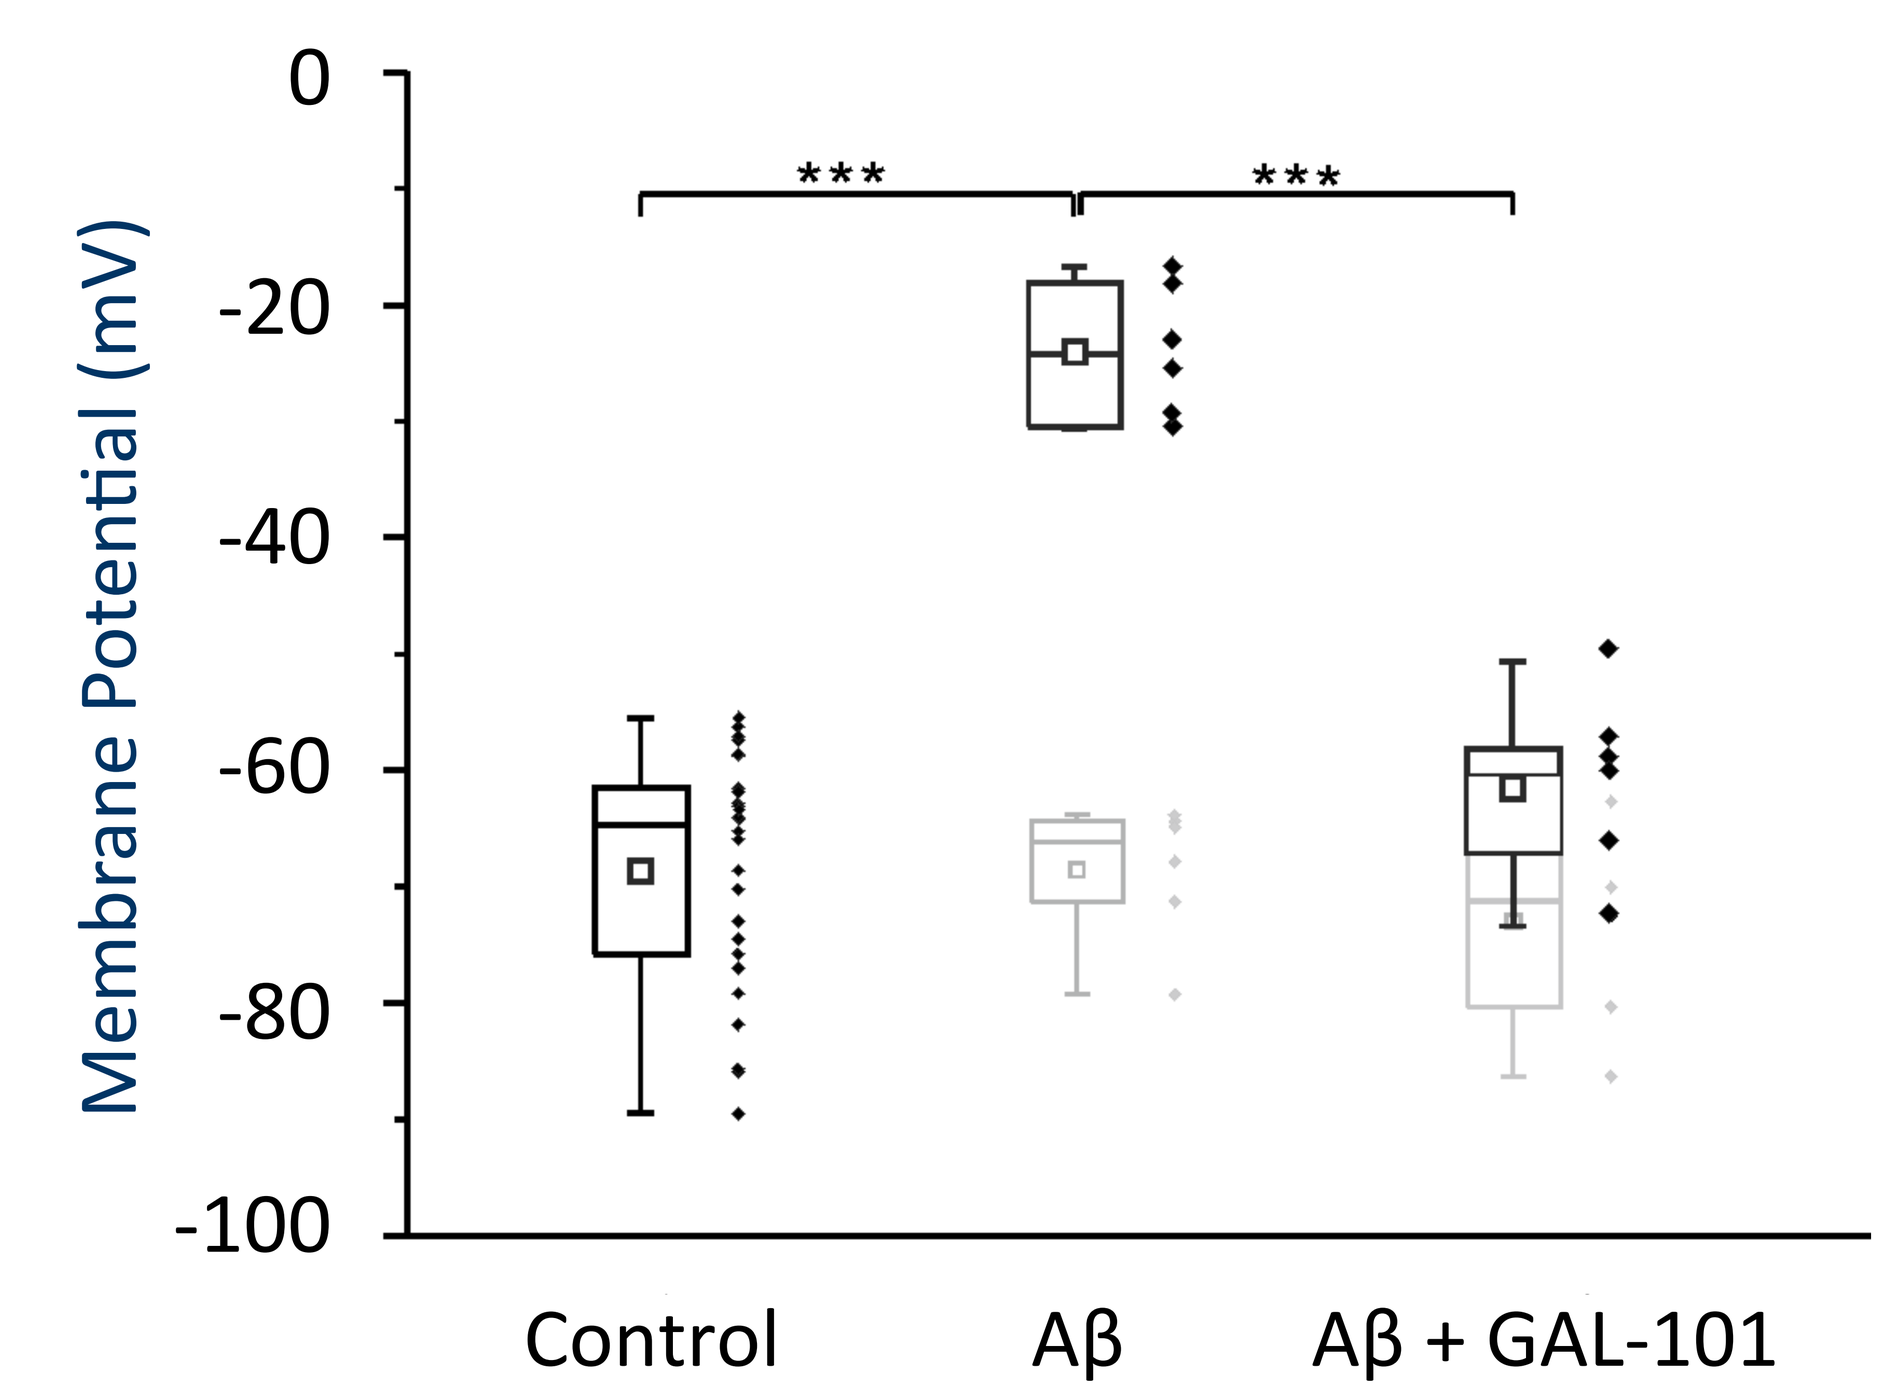
Membrane Potential (mV)

Control
Aβ
Aβ + GAL-101

Conductance in lipid bilayers

GAL-101 prevents formation of Aβ pores / channels in lipid bilayers

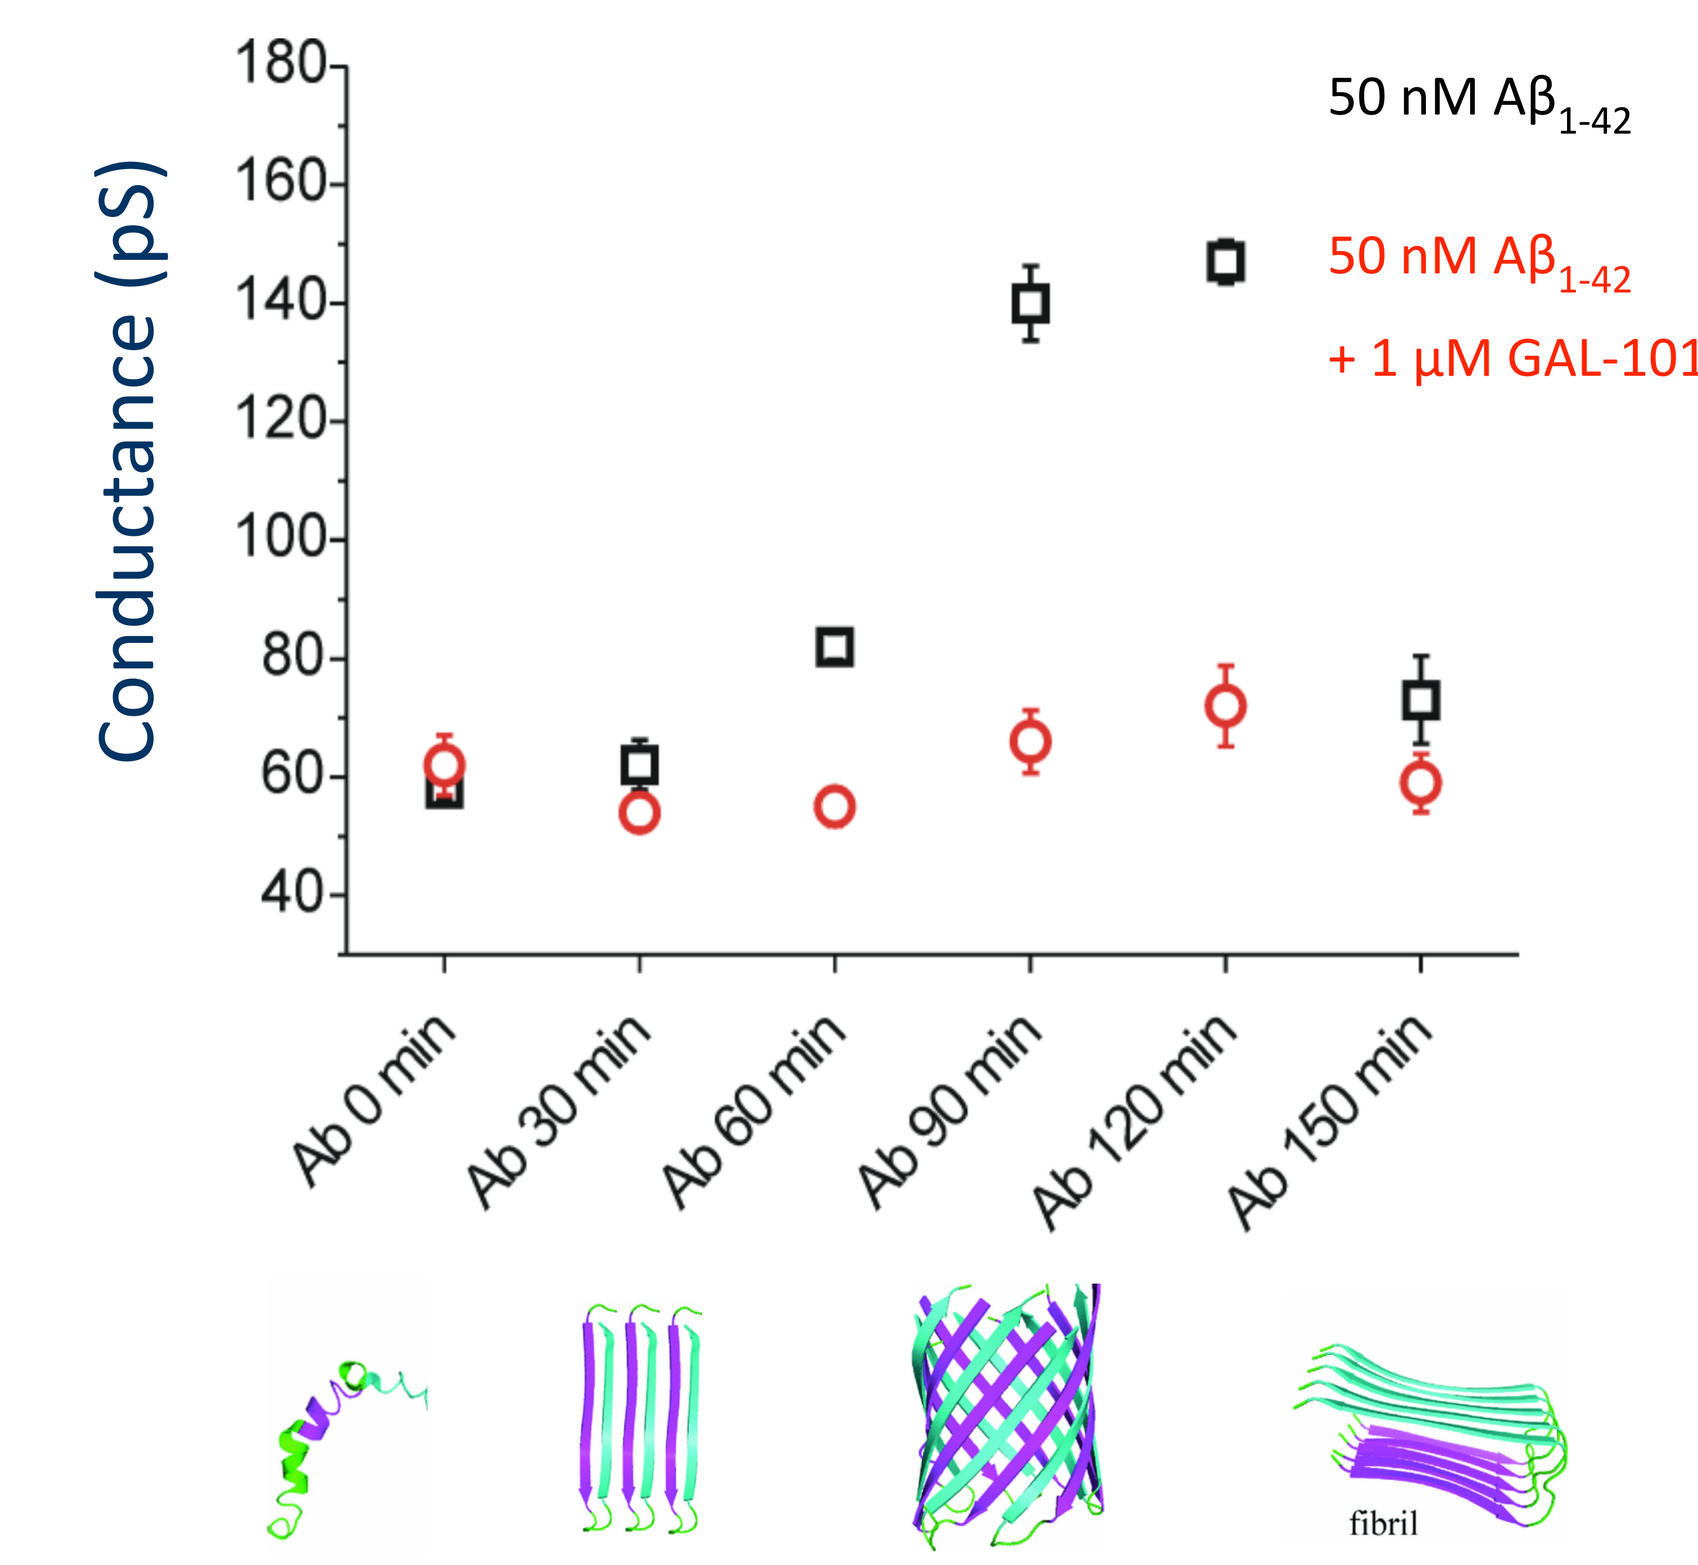
Conductance (pS)
50 nM $Aβ_{1-42}$
50 nM $Aβ_{1-42}$ + 1 µM GAL-101
Ab 0 min
Ab 30 min
Ab 60 min
Ab 90 min
Ab 120 min
Ab 150 min
fibril

Resting membrane potential measurements in Retinal Pigment Epilthelial Cells (RPEs)

Resting membrane potential measurements in Retinal Pigment Epithelial Cells (RPEs).

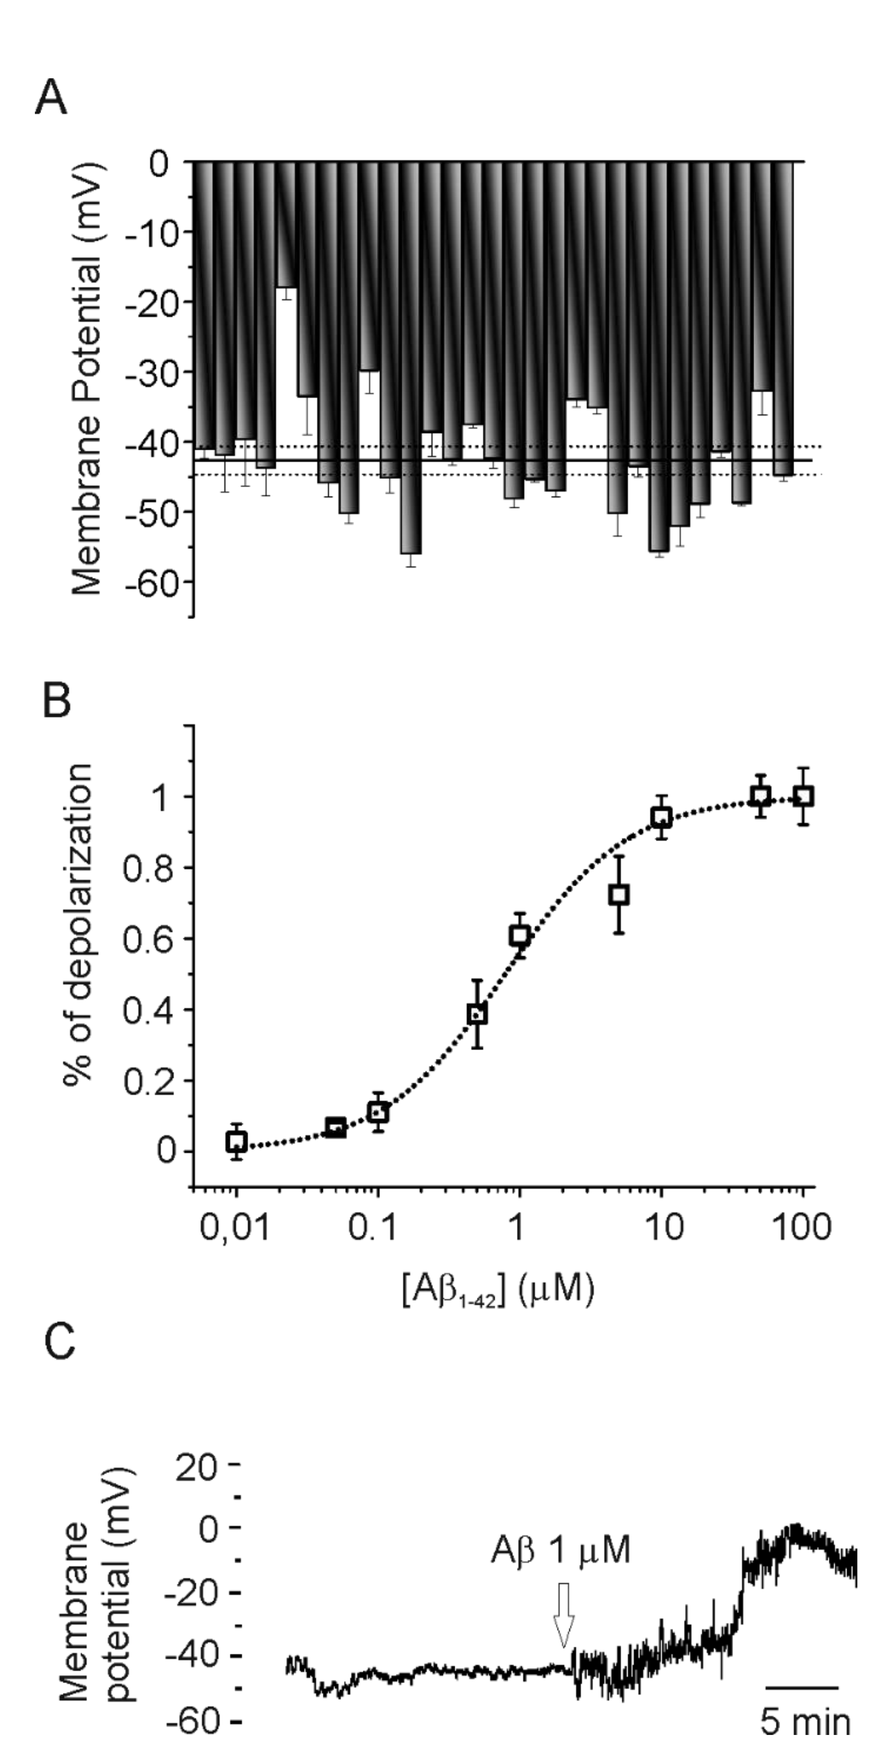
A
Membrane Potential (mV)
B
% of depolarization
$[Aβ_{1-42}]$ (µM)
C
Membrane potential (mV)
Aβ 1 µM
5 min

GAL-101 prevents RPEs membrane depolarization induced by $Aβ_{1-42}$

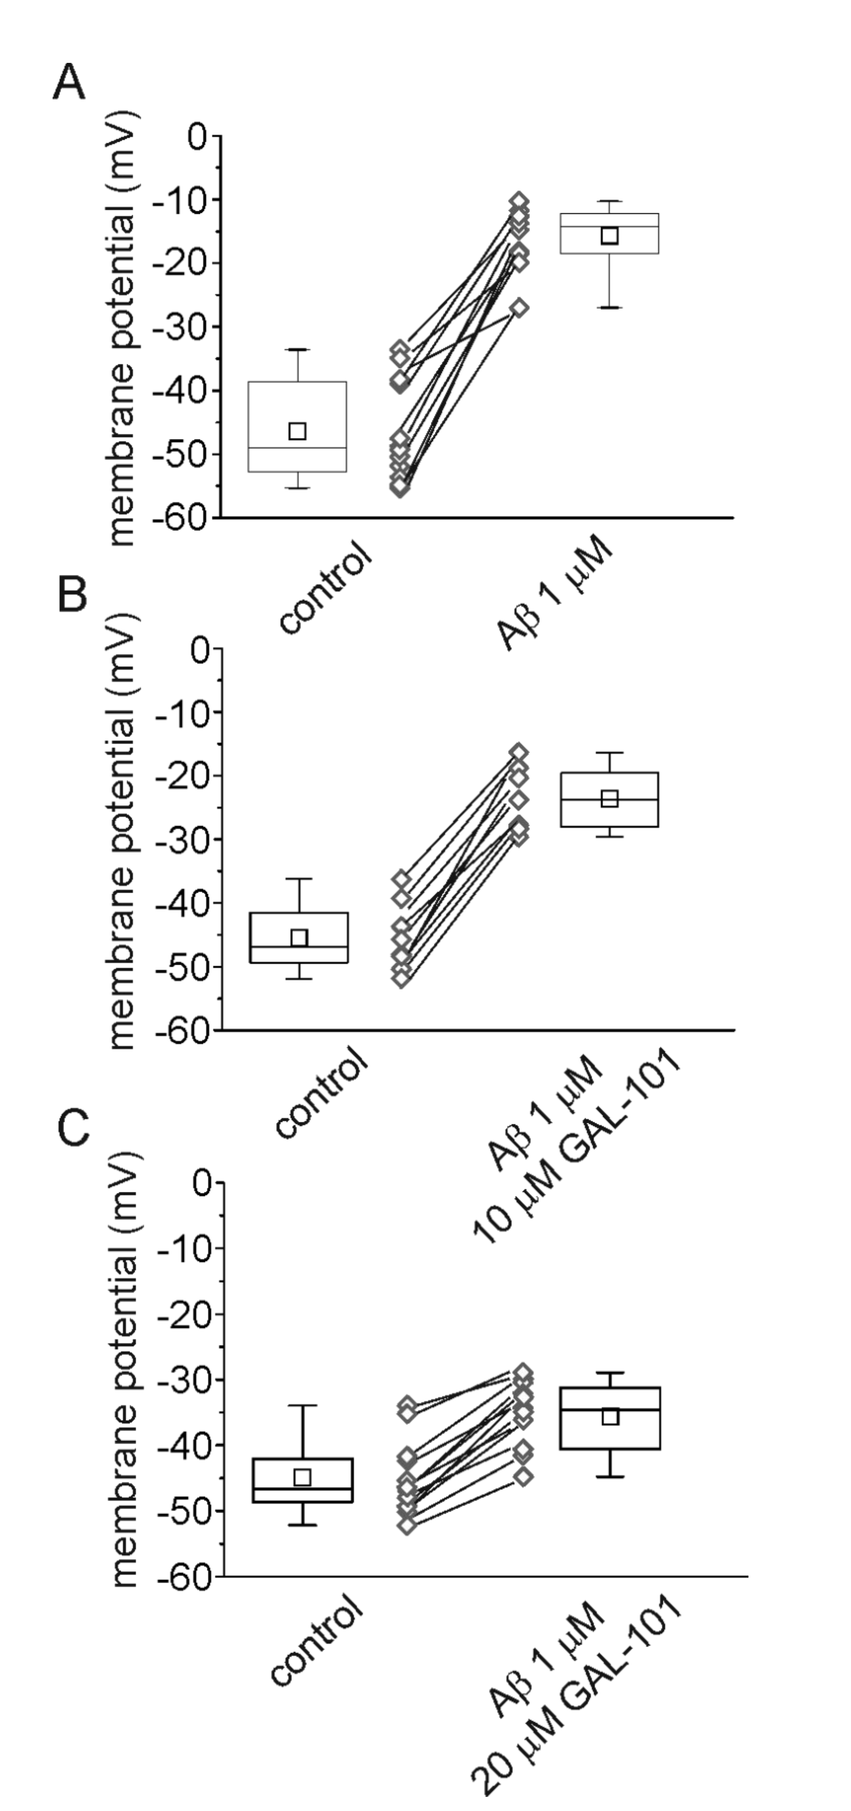
A
membrane potential (mV)
control
Aβ 1 µM
B
Aβ 1 µM 10 µM GAL-101
C
Aβ 1 µM 20 µM GAL-101

Efficacy and Mechanism of Action (MoA) of GAL-101 supported by a variety of different techniques / models

Efficacy and Mechanism of Action (MoA) of GAL-101

1. GAL-101 binds selectively to misfolded Aβ monomers
2. Clusters have been visualized using atomic force microscopy
3. Reduction of deposited β-sheet Aβ demonstrated in animal models
4. Detoxification demonstrated by functional tests of GAL-101 in cell culture
5. Dilution experiments demonstrated detoxifying activity of clusters

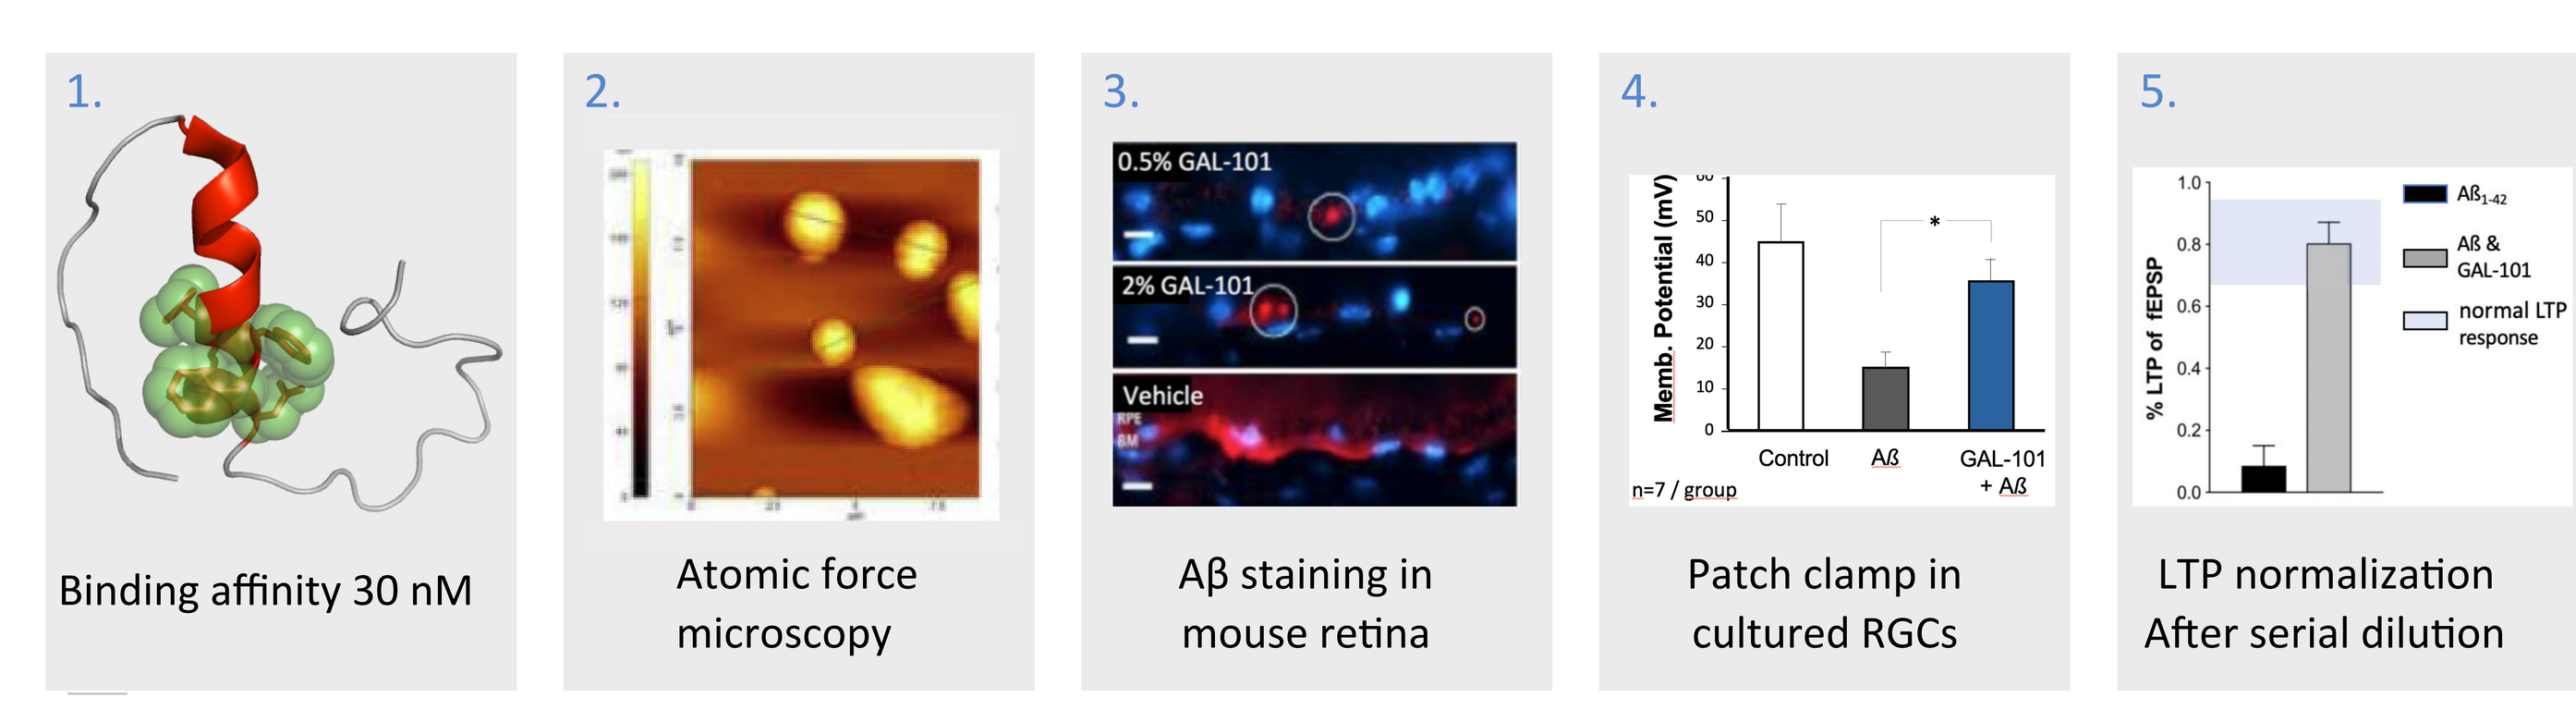
1.
Binding affinity 30 nM
2.
Atomic force microscopy
3.
0.5% GAL-101
2% GAL-101
Vehicle
Aβ staining in mouse retina
4.
Memb. Potential (mV)
Control
Aβ
GAL-101 + Aβ
n=7 / group
Patch clamp in cultured RGCs
5.
% LTP of fEPSP
normal LTP response
LTP normalization After serial dilution
6

Efficacy and Mechanism of Action (MoA) of GAL-101

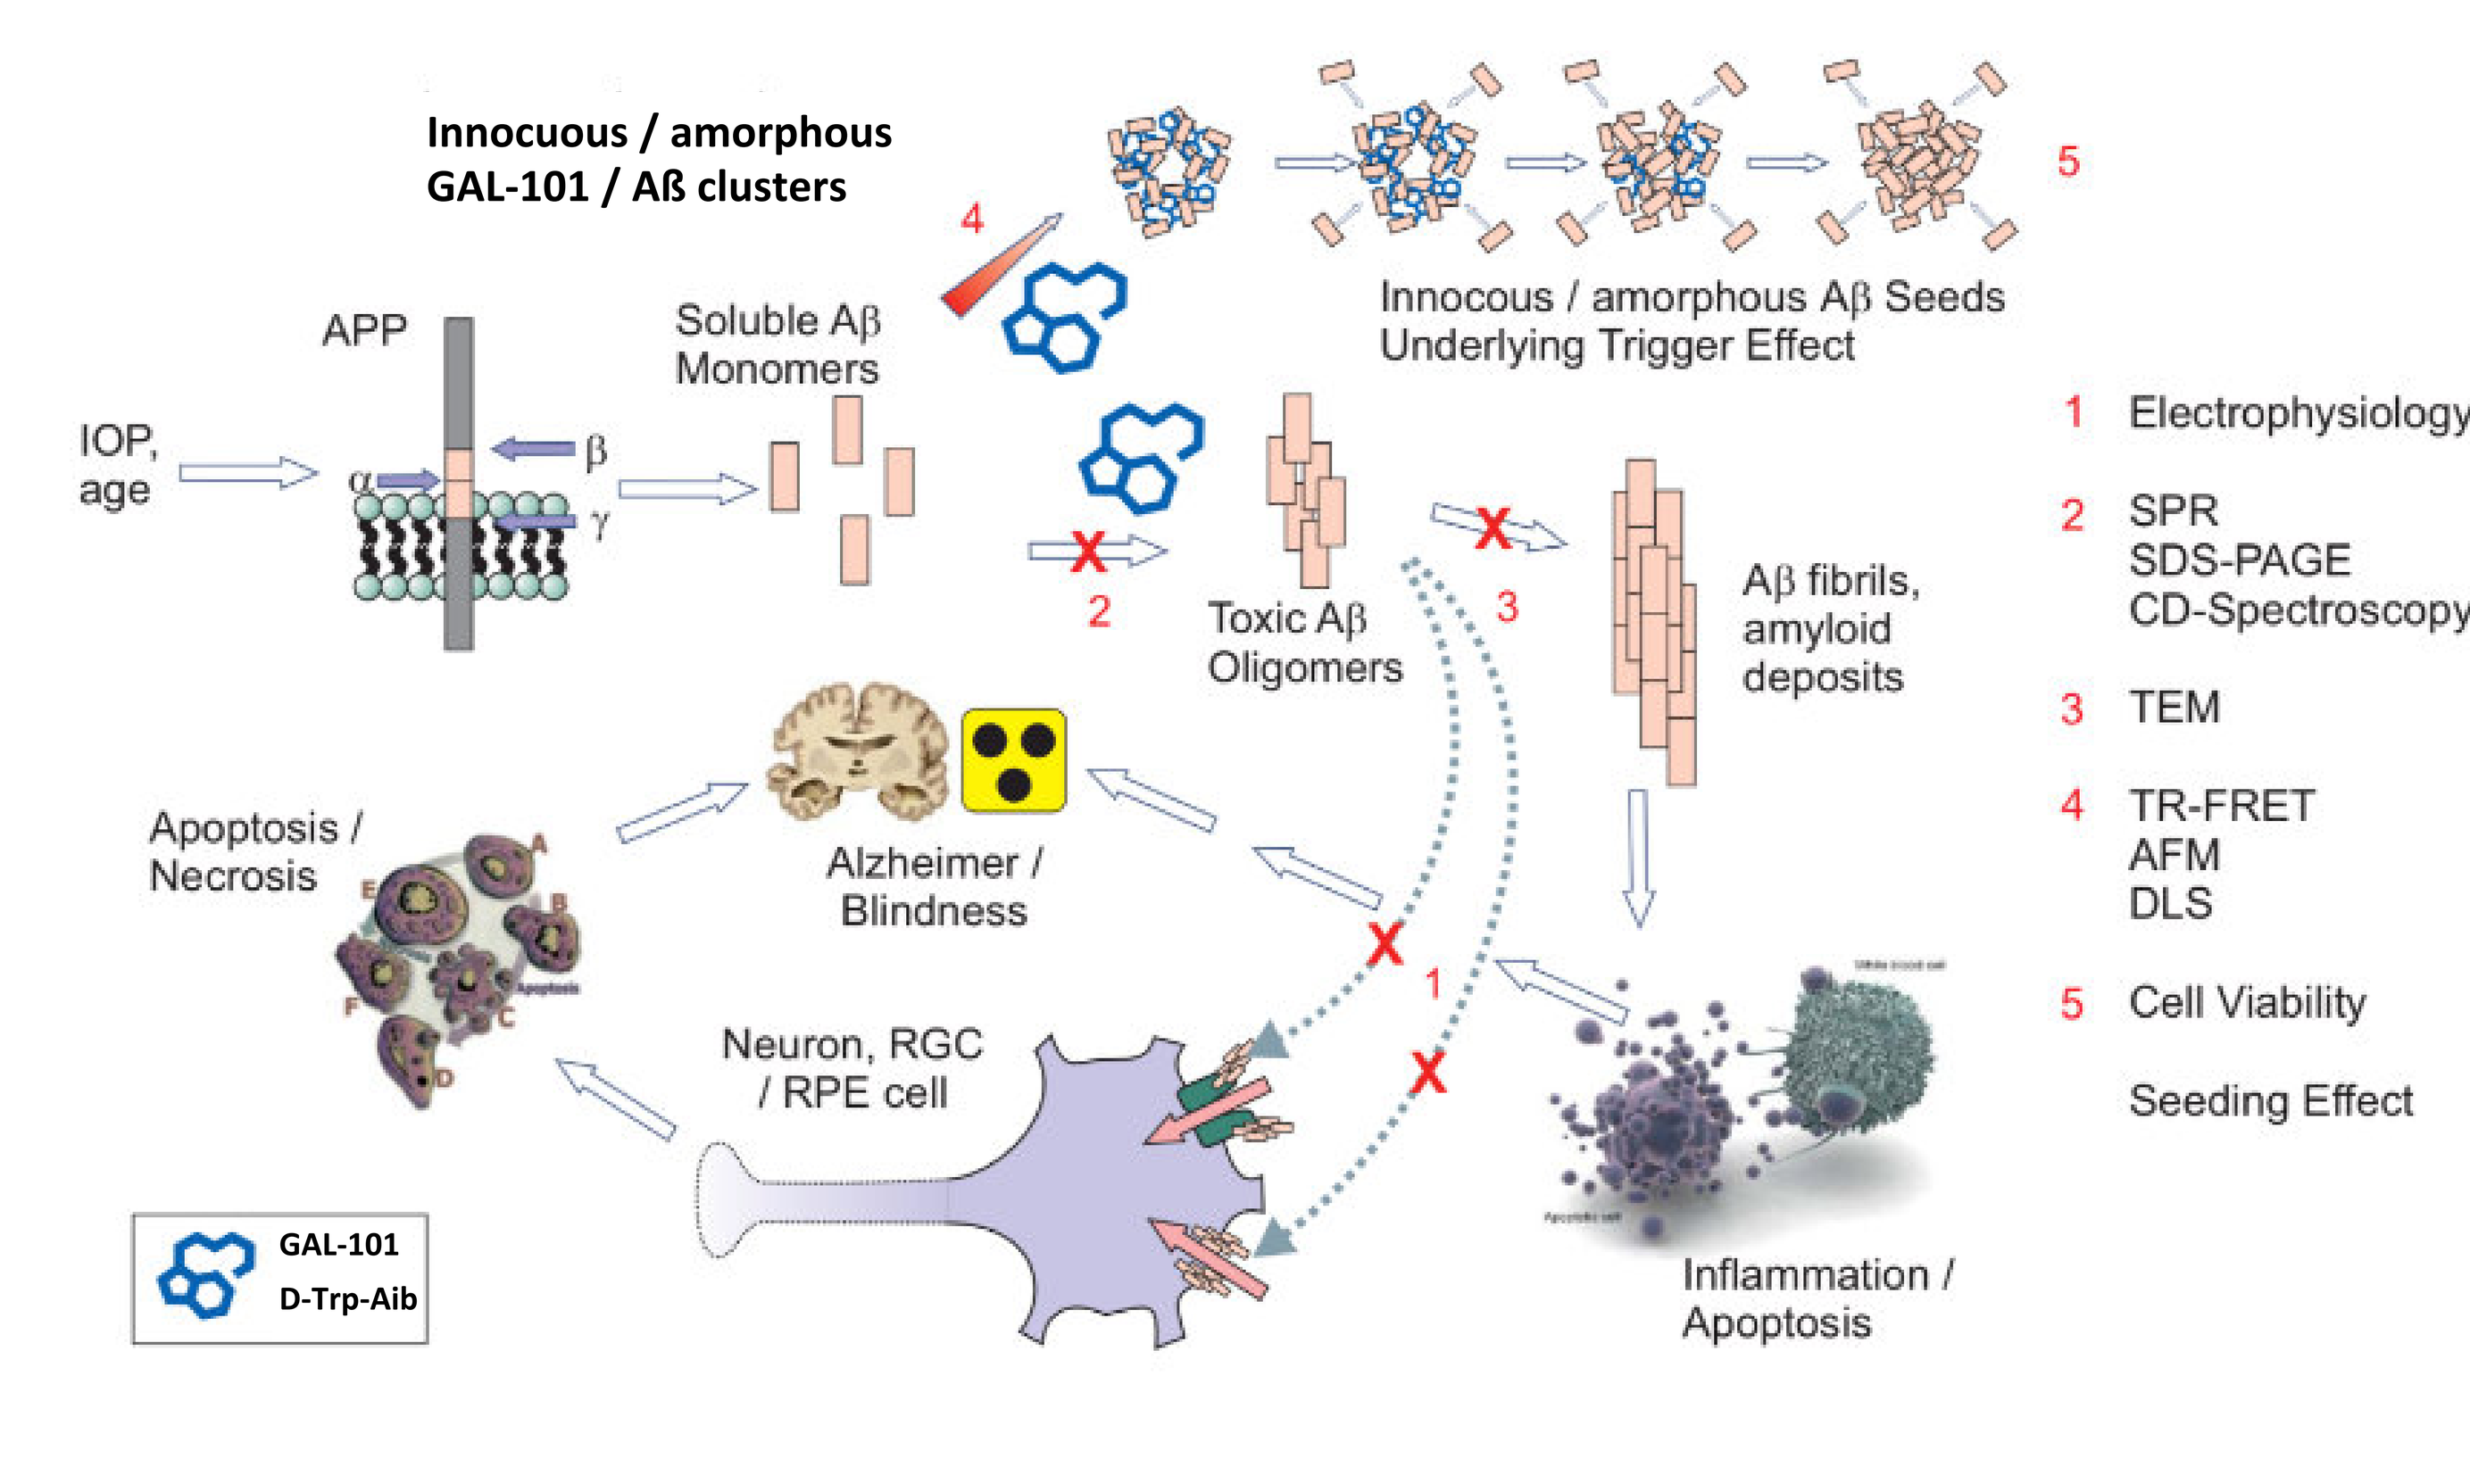
Innocuous / amorphous GAL-101 / Aß clusters
Innocous / amorphous Aβ Seeds Underlying Trigger Effect
APP
IOP, age
Soluble Aβ Monomers
Toxic Aβ Oligomers
Aβ fibrils, amyloid deposits
Apoptosis / Necrosis
Alzheimer / Blindness
Neuron, RGC / RPE cell
Inflammation / Apoptosis
GAL-101 D-Trp-Aib
1 Electrophysiology
2 SPR SDS-PAGE CD-Spectroscopy
3 TEM
4 TR-FRET AFM DLS
5 Cell Viability
Seeding Effect