Abstract 50
PB-001

SY-1365, a selective CDK7 inhibitor, enhances carboplatin activity in ovarian cancer cell lines and xenografts, and inhibits effectors of homologous recombination repair

Liv Johannessen, Shanhu Hu, Nan Ke, Nisha Rajagopal, David Orlando, Sydney Alnemy, Anthony D'Ippolito, John Carulli, Emmanuelle di Tomaso, Graeme Hodgson
Syros Pharmaceuticals, 620 Memorial Drive, Cambridge MA 02139

Introduction

- Homologous Recombination Deficiency (HRD) forms the basis for synthetic lethal interactions with agents that directly cause DNA damage (e.g. carboplatin) or agents that inhibit DNA repair (e.g. PARP inhibitors).
- HRD is caused by alterations in effectors of homologous recombination repair (HRR), such as mutation of BRCA1/2 or ATM, and ~50% of high-grade serous ovarian cancers have alterations in the HRD pathway.
- CDK7 is key regulator of transcription and cell cycle progression and has been implicated in the regulation of DNA damage repair responses in a variety of model systems.
- SY-1365, a covalent and selective inhibitor of CDK7 in Phase 1 clinical development, has previously been shown to induce cytotoxicity in ovarian cancer (OC) cell lines *in vitro* and induce tumor growth inhibition, including complete regressions, in OC patient-derived xenograft models.
- Here we report on the activity of SY-1365 in combination with the DNA damaging agent carboplatin (CP) in OC preclinical models *in vitro* and *in vivo.*

Homologous recombination deficiency (HRD) forms the basis for synthetic lethality with DNA damaging agents

HRD is caused by alterations in effectors of homologous recombination repair (HRR)

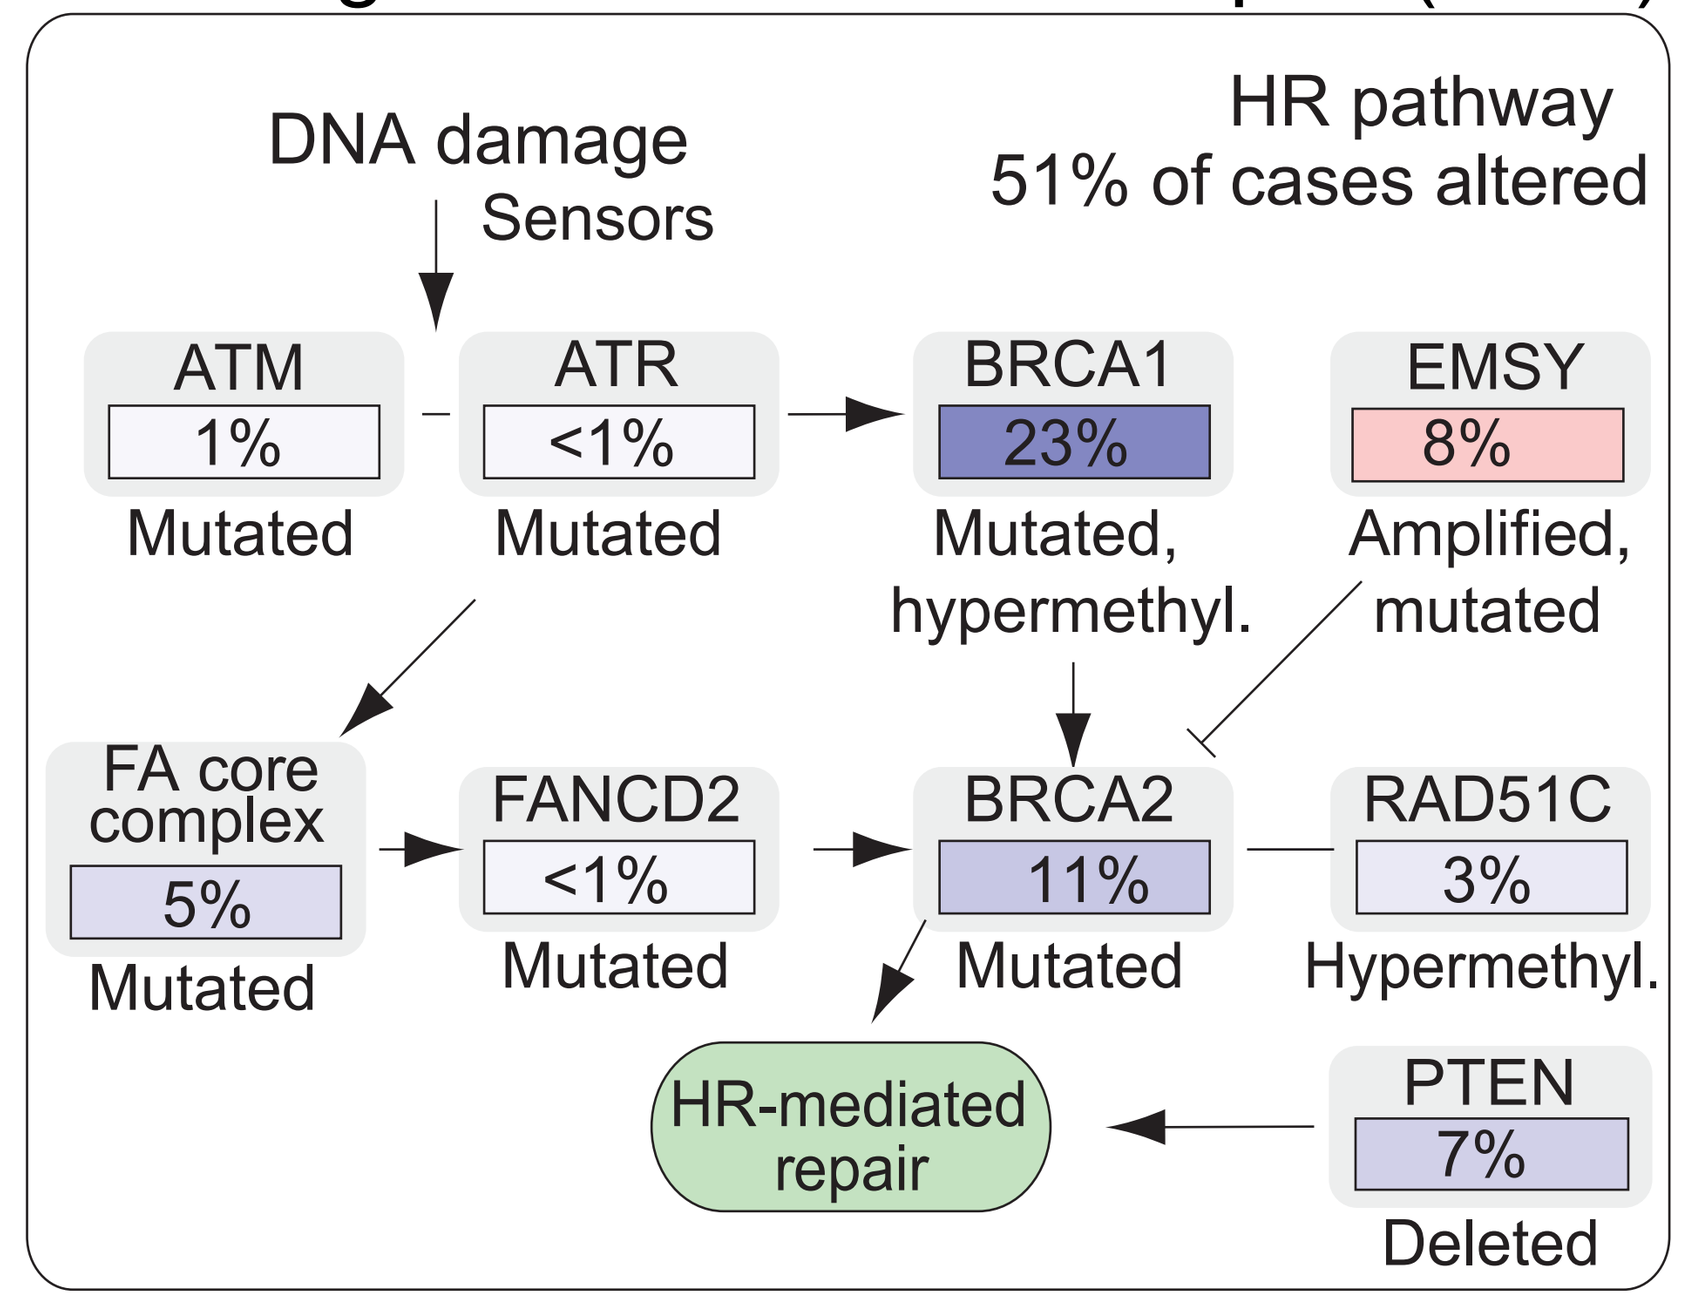
DNA damage
Sensors
HR pathway
51% of cases altered
ATM
1%
Mutated
ATR
<1%
Mutated
BRCA1
23%
Mutated, hypermethyl.
EMSY
8%
Amplified, mutated
FA core complex
5%
Mutated
FANCD2
<1%
Mutated
BRCA2
11%
Mutated
RAD51C
3%
Hypermethyl.
HR-mediated repair
PTEN
7%
Deleted

(TCGA, Nature 2011

Proposed model of SY-1365 synthetic lethal interaction with CP

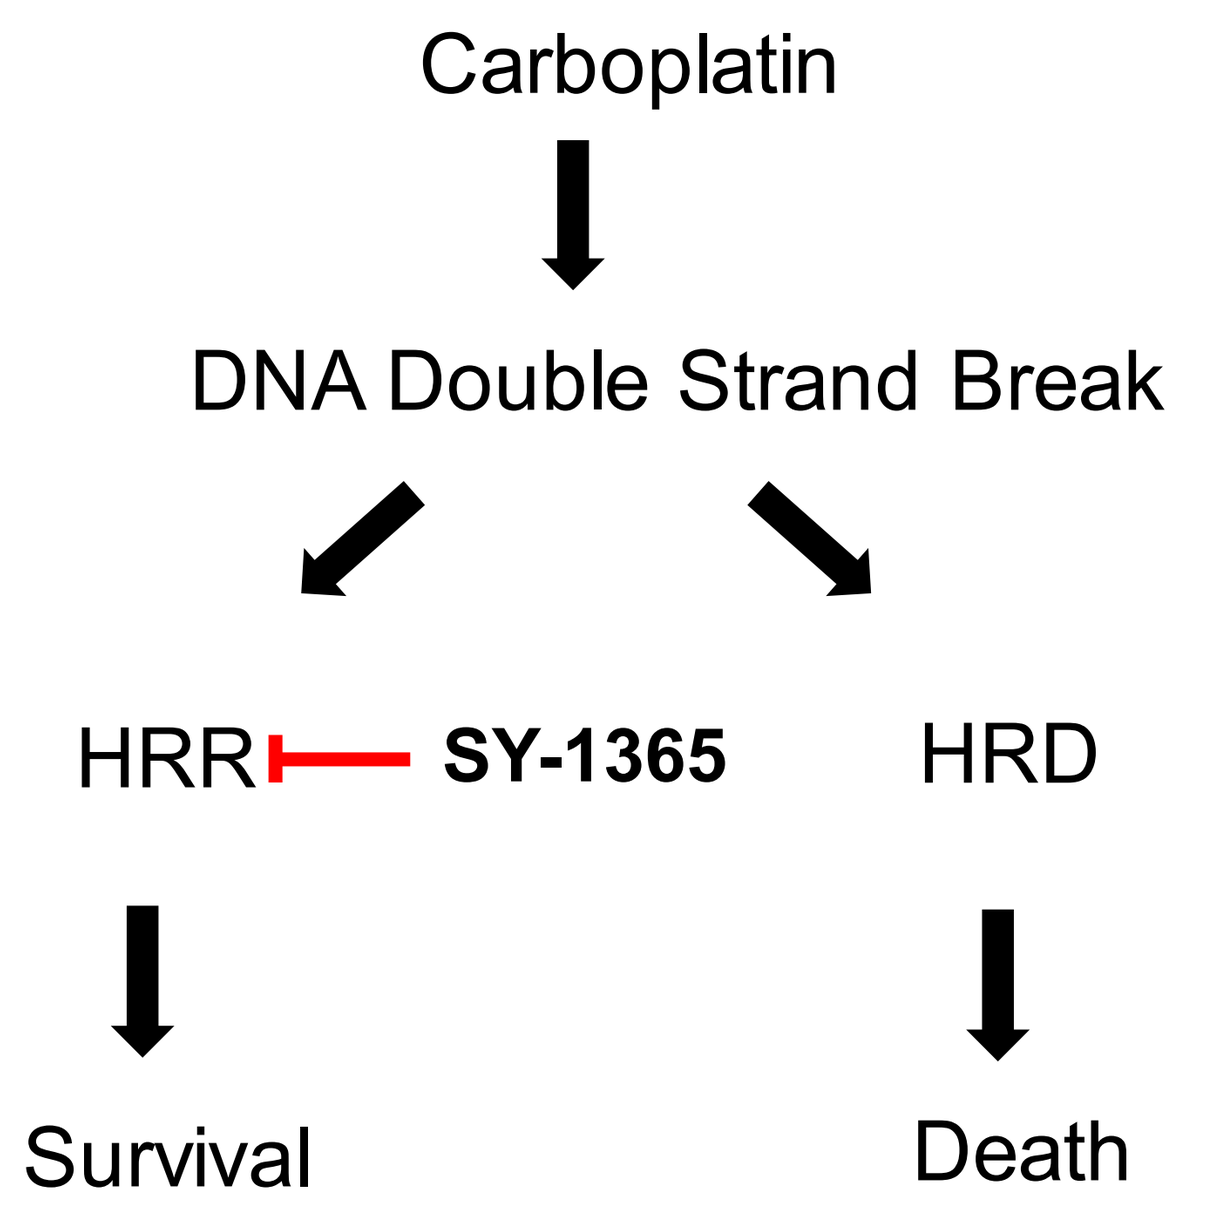
Carboplatin
DNA Double Strand Break
HRR
SY-1365
HRD
Survival
Death

A subset of HGSOC exhibit HRD, and exhibit superior response to platinum-agents. HRD in HGSOC is caused by alterations in effectors of HRR (commonly BRCA1/2 mutations), thus are unable to repair the DNA damage caused by platinum agents. This supports the therapeutic rationale for induction of HRD in HGSOC patients to allow for increased duration and depth of response to platinum agents. SY-1365 shows synergy with CP in OC models both *in vitro* and *in vivo*. Furthermore, SY-1365 transcriptionally downregulates key effectors of HRR in OC and CP-induced HRR, as measured by nuclear RAD51 foci formation. These results suggest that SY-1365 treatment may induce an HRD-like state in ovarian cancer models, supporting the rationale for combination of SY-1365 with carboplatin in HGSOC.

SY-1365 is synergistic with carboplatin in ovarian cancer cell lines and enhances carboplatin activity in ovarian cancer xenografts

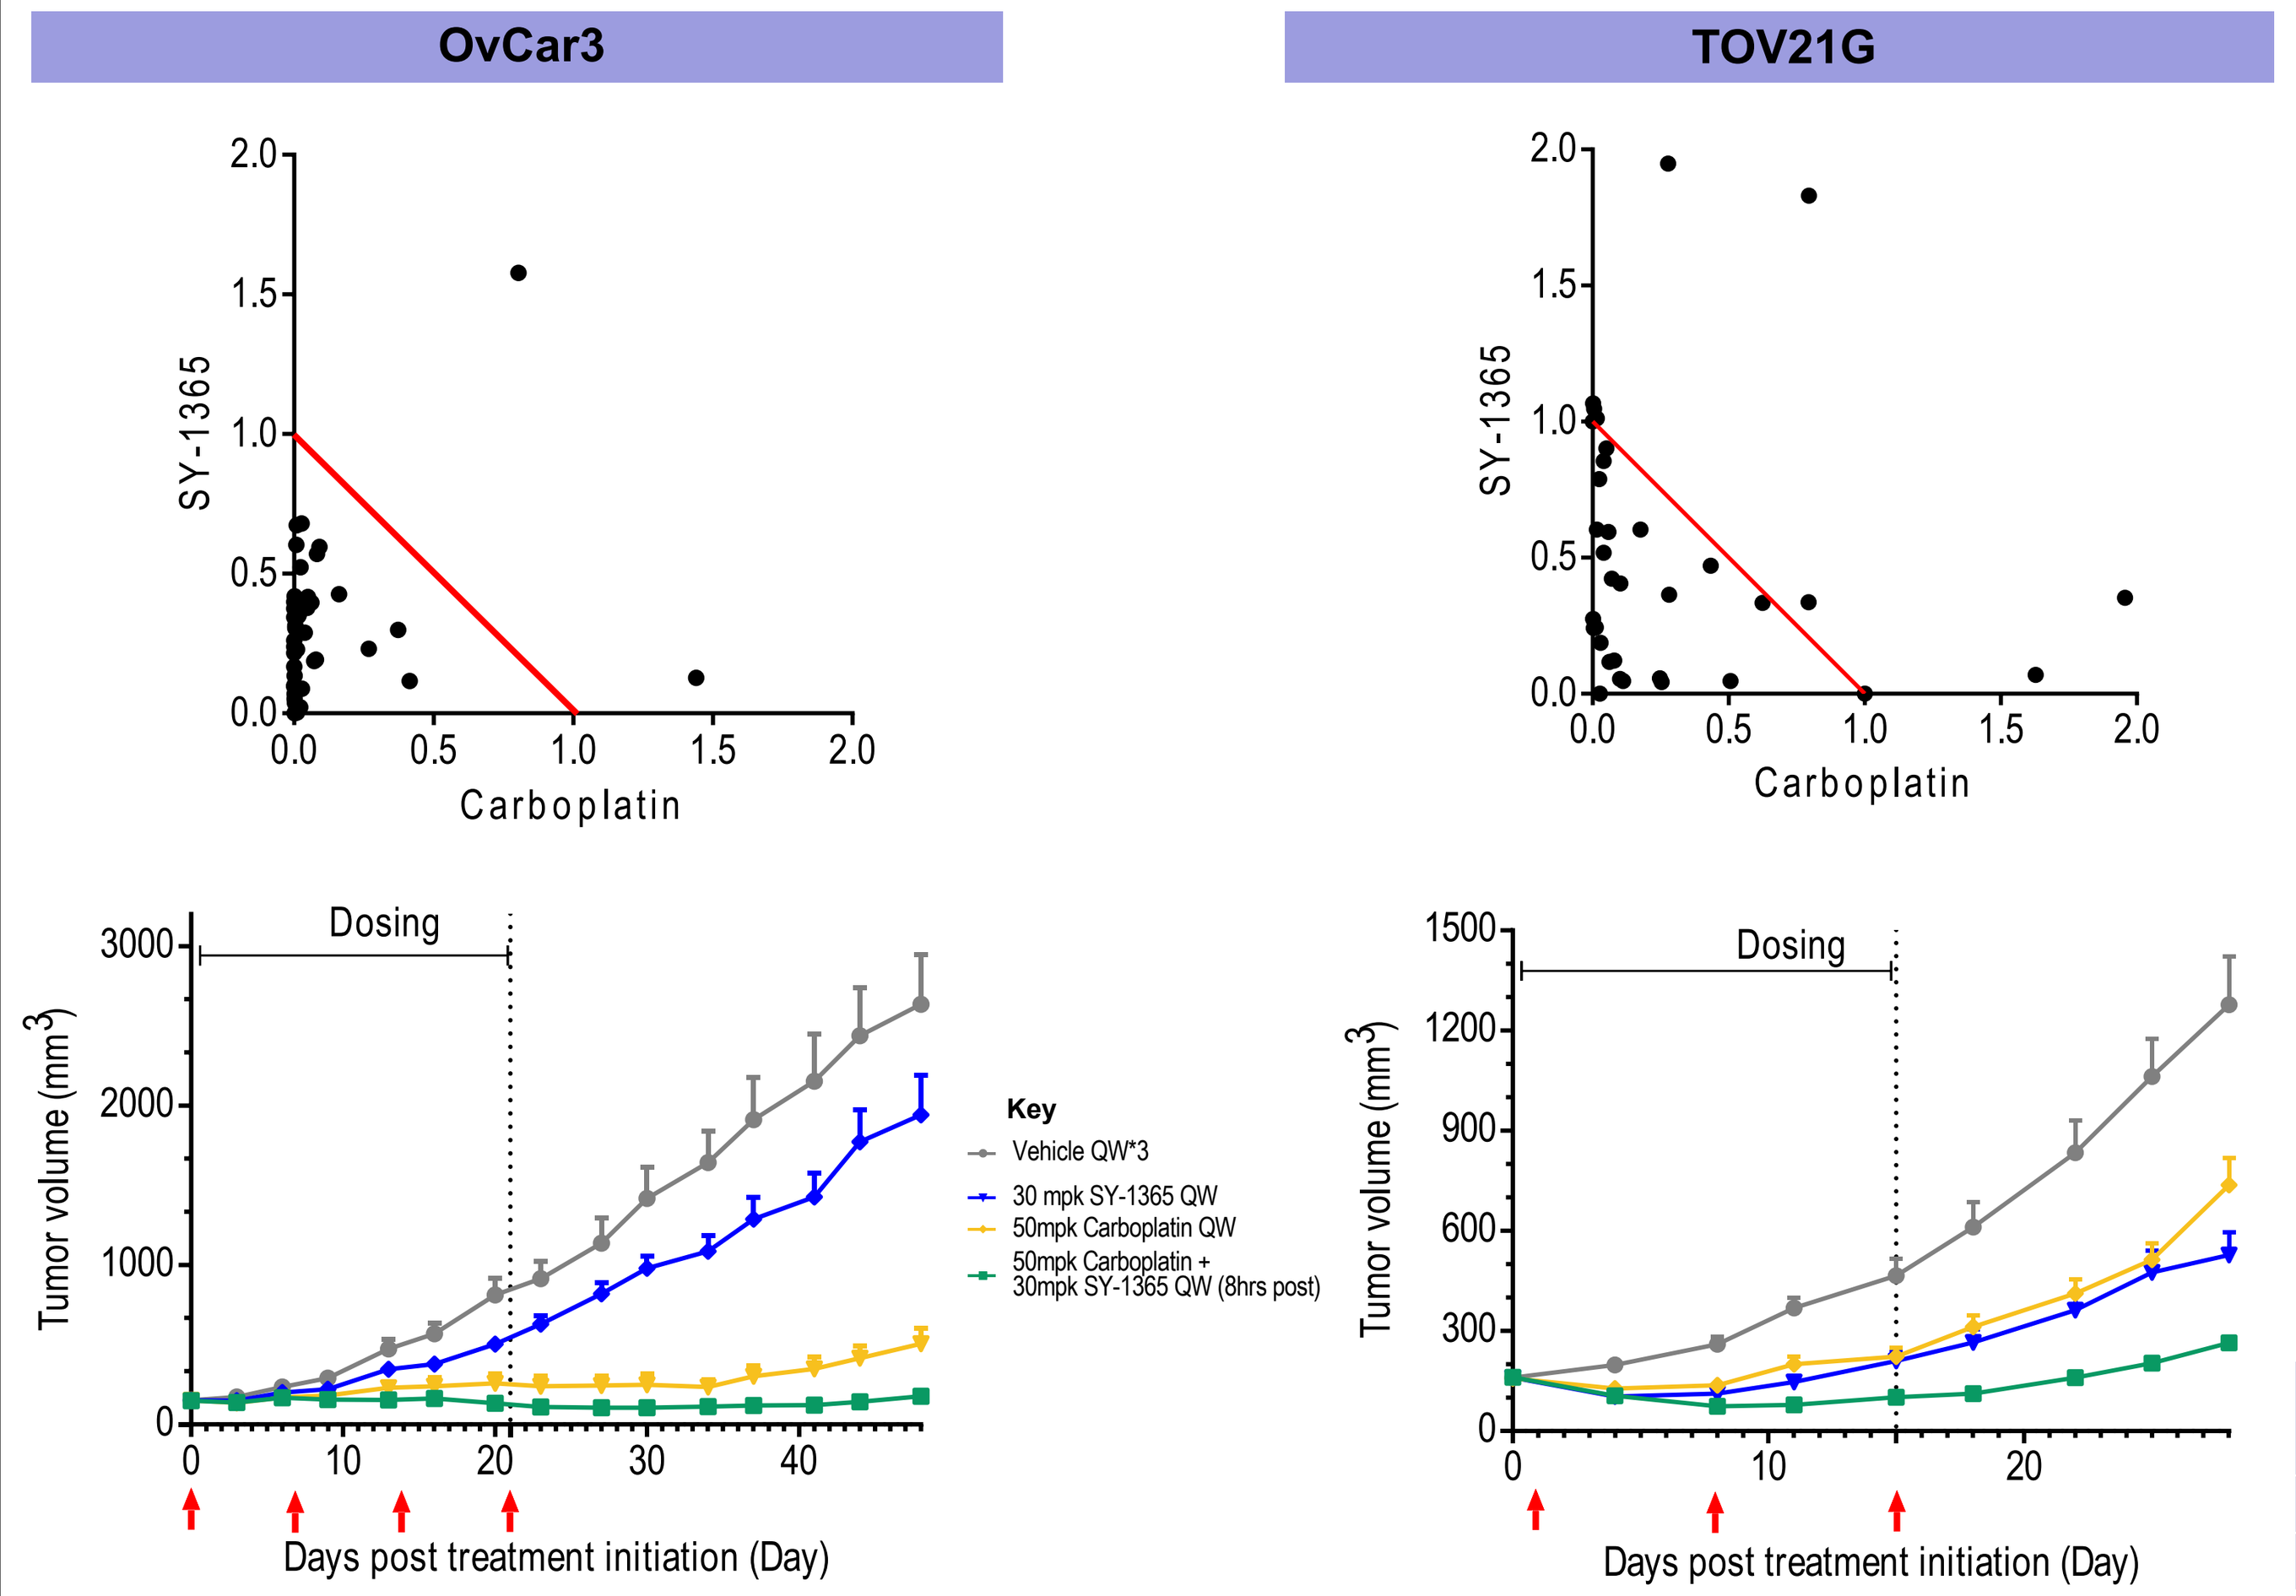
OvCar3
TOV21G
SY-1365
Carboplatin
Dosing
Tumor volume (mm3)
Days post treatment initiation (Day)
Key
Vehicle QW*3
30 mpk SY-1365 QW
50mpk Carboplatin QW
50mpk Carboplatin + 30mpk SY-1365 QW (8hrs post)

In vitro (top): Dose-response curves were generated in OC cell lines (TOV21G, OVCAR3) *in vitro* with SY-1365 combination with CP; isobolograms and synergy were calculated using the Chou-Talalay method.

In vivo (bottom): Growth of ovarian CDX models (OvCar3 Crown Bioscience and TOV21G ChemPartner) with once weekly (QW) dosing for 3 or 4 weeks (red arrows) (CP 50mpk, IP; SY-1365 30mpk, IV. *Note: SY-1365 MTD: 40mpk, IV, BIW.* Curves represent average tumor growth of n=10 treated mice.

Xenograft	Tumor Growth Inhibition (%)		
	CP	SY-1365	CP + SY-1365
OVCAR3	88	37	105
TOV21G	79	84	120

SY-1365 transcriptionally downregulates key mediators of the homologous recombination repair pathway in ovarian cancer cell lines and xenografts

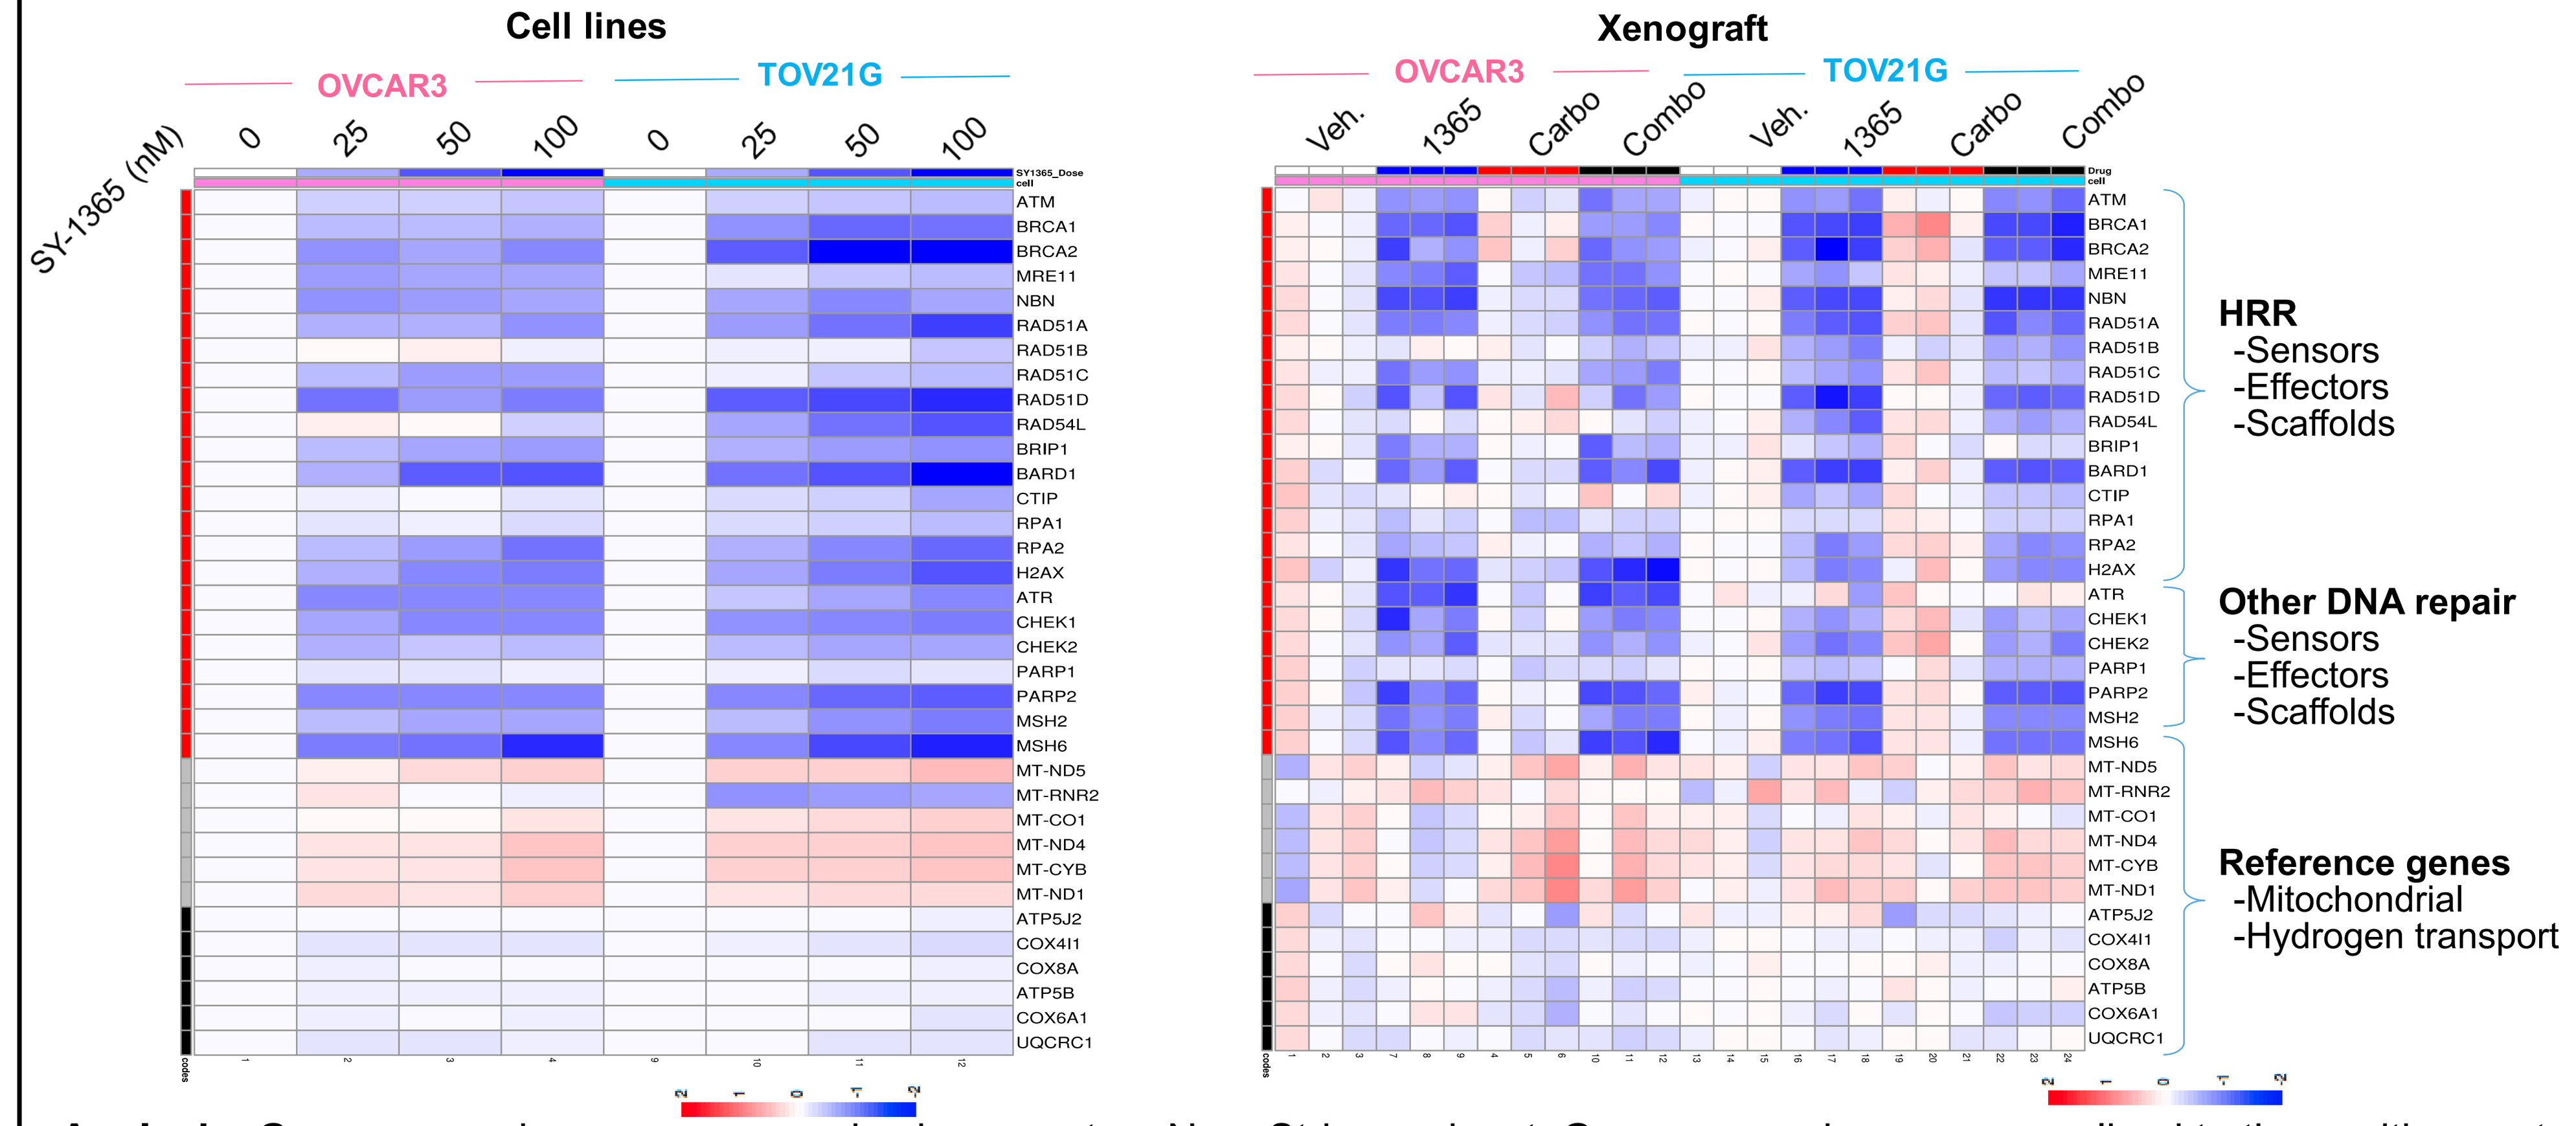
Cell lines
Xenograft
OVCAR3
TOV21G
SY-1365 (nM)
0
25
50
100
Veh.
1365
Carbo
Combo
ATM
BRCA1
BRCA2
MRE11
NBN
RAD51A
RAD51B
RAD51C
RAD51D
RAD54L
BRIP1
BARD1
CTIP
RPA1
RPA2
H2AX
ATR
CHEK1
CHEK2
PARP1
PARP2
MSH2
MSH6
MT-ND5
MT-RNR2
MT-CO1
MT-ND4
MT-CYB
MT-ND1
ATP5J2
COX4I1
COX8A
ATP5B
COX6A1
UQCRC1
HRR
-Sensors
-Effectors
-Scaffolds
Other DNA repair
-Sensors
-Effectors
-Scaffolds
Reference genes
-Mitochondrial
-Hydrogen transport

Analysis: Gene expression was measured using a custom NanoString codeset. Gene expression was normalized to the positive control spike-ins and reference genes. Log2 fold changes were calculated against the average matched vehicle signal. ***In vitro,*** samples collected 6h post-dose. ***In vivo,*** SY-1365, 30mpk QW; Carboplatin, 50mpk QW; combination, single agent doses, SY-1365 dosed 8h after carboplatin; tumor samples collected 6h after second dose of SY-1365.

SY-1365 inhibits RAD51 nuclear foci formation following carboplatin-induced DNA damage in ovarian cancer cell lines in vitro

***In vitro* imaging (representative images, right):** OVCAR3 or TOV21G cells were treated with either CP, SY-1365 or the combination for 48 hours. Cells were visualized using immunofluorescence microscopy to determine the levels of DNA damage (via nuclear pH2A.X) and the cells' ability to repair that damage using HRR (via RAD51 nuclear foci). CP alone induces both an increase in pH2A.x, and nuclear RAD51 foci.

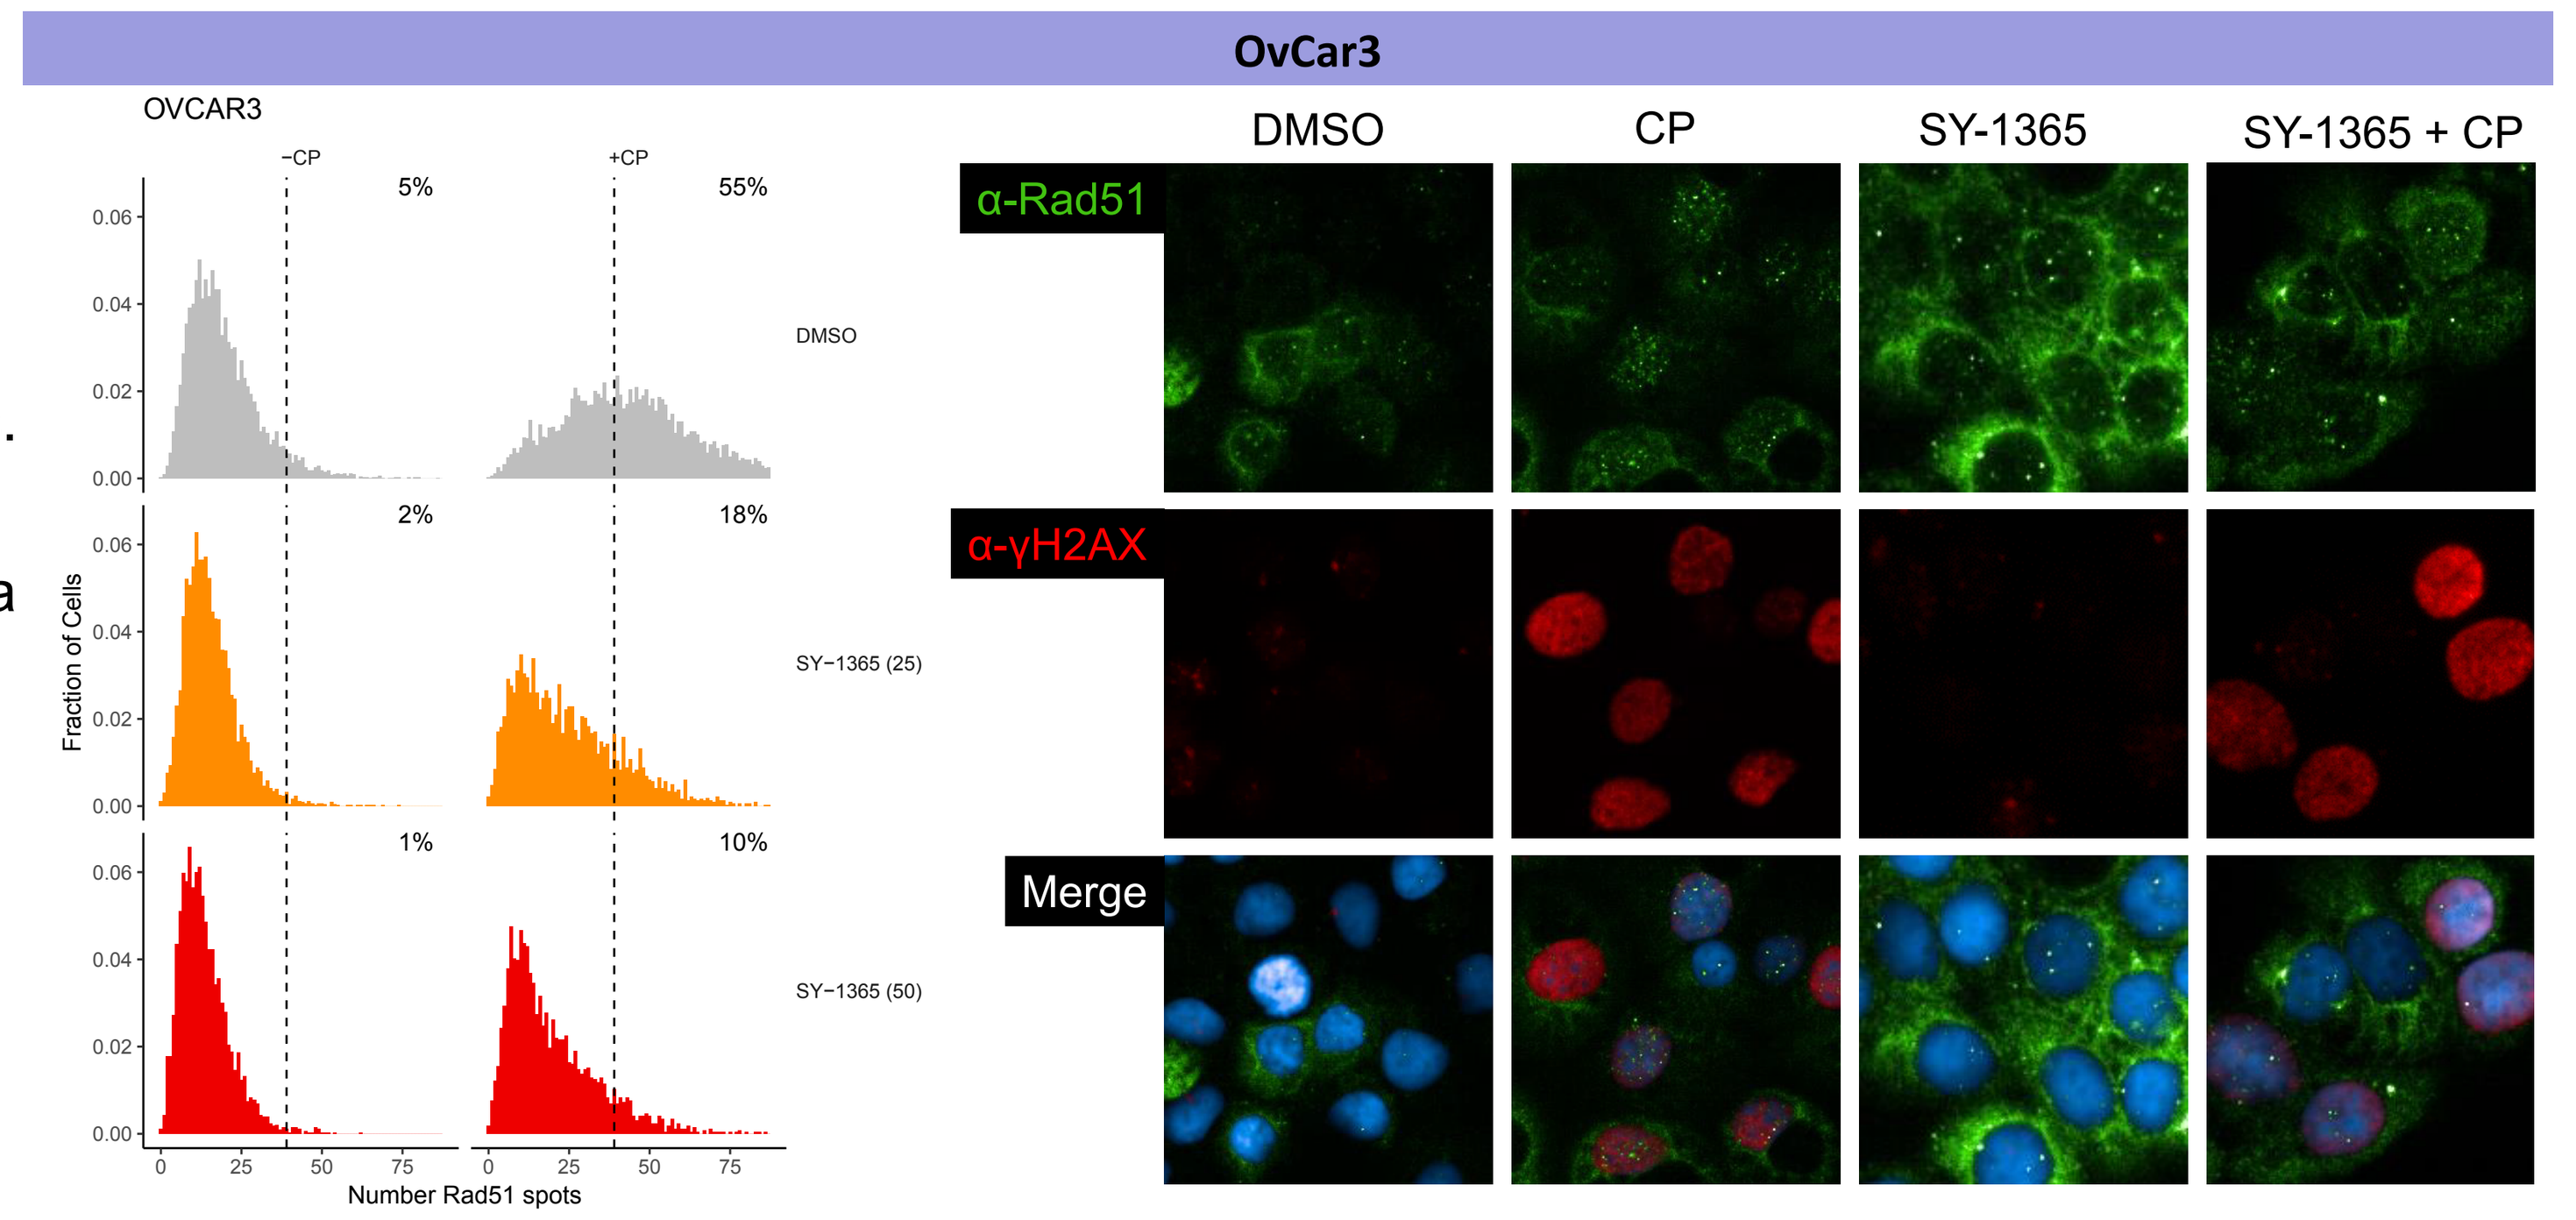
OvCar3
OVCAR3
−CP
+CP
5%
55%
2%
18%
1%
10%
DMSO
SY−1365 (25)
SY−1365 (50)
Fraction of Cells
Number Rad51 spots
DMSO
CP
SY-1365
SY-1365 + CP
α-Rad51
α-γH2AX
Merge

Analysis (histograms, left): Single-cell image analysis was used to quantify the number of RAD51 nuclear foci. As a reference, we set a cutoff at the 95th percentile of RAD51 spots in the DMSO condition (dashed line, left figures). RAD51 foci increase upon treatment with CP. When combined with SY-1365, however, the pH2A.x was sustained, but the presence of RAD51 foci was significantly decreased in both cell lines.

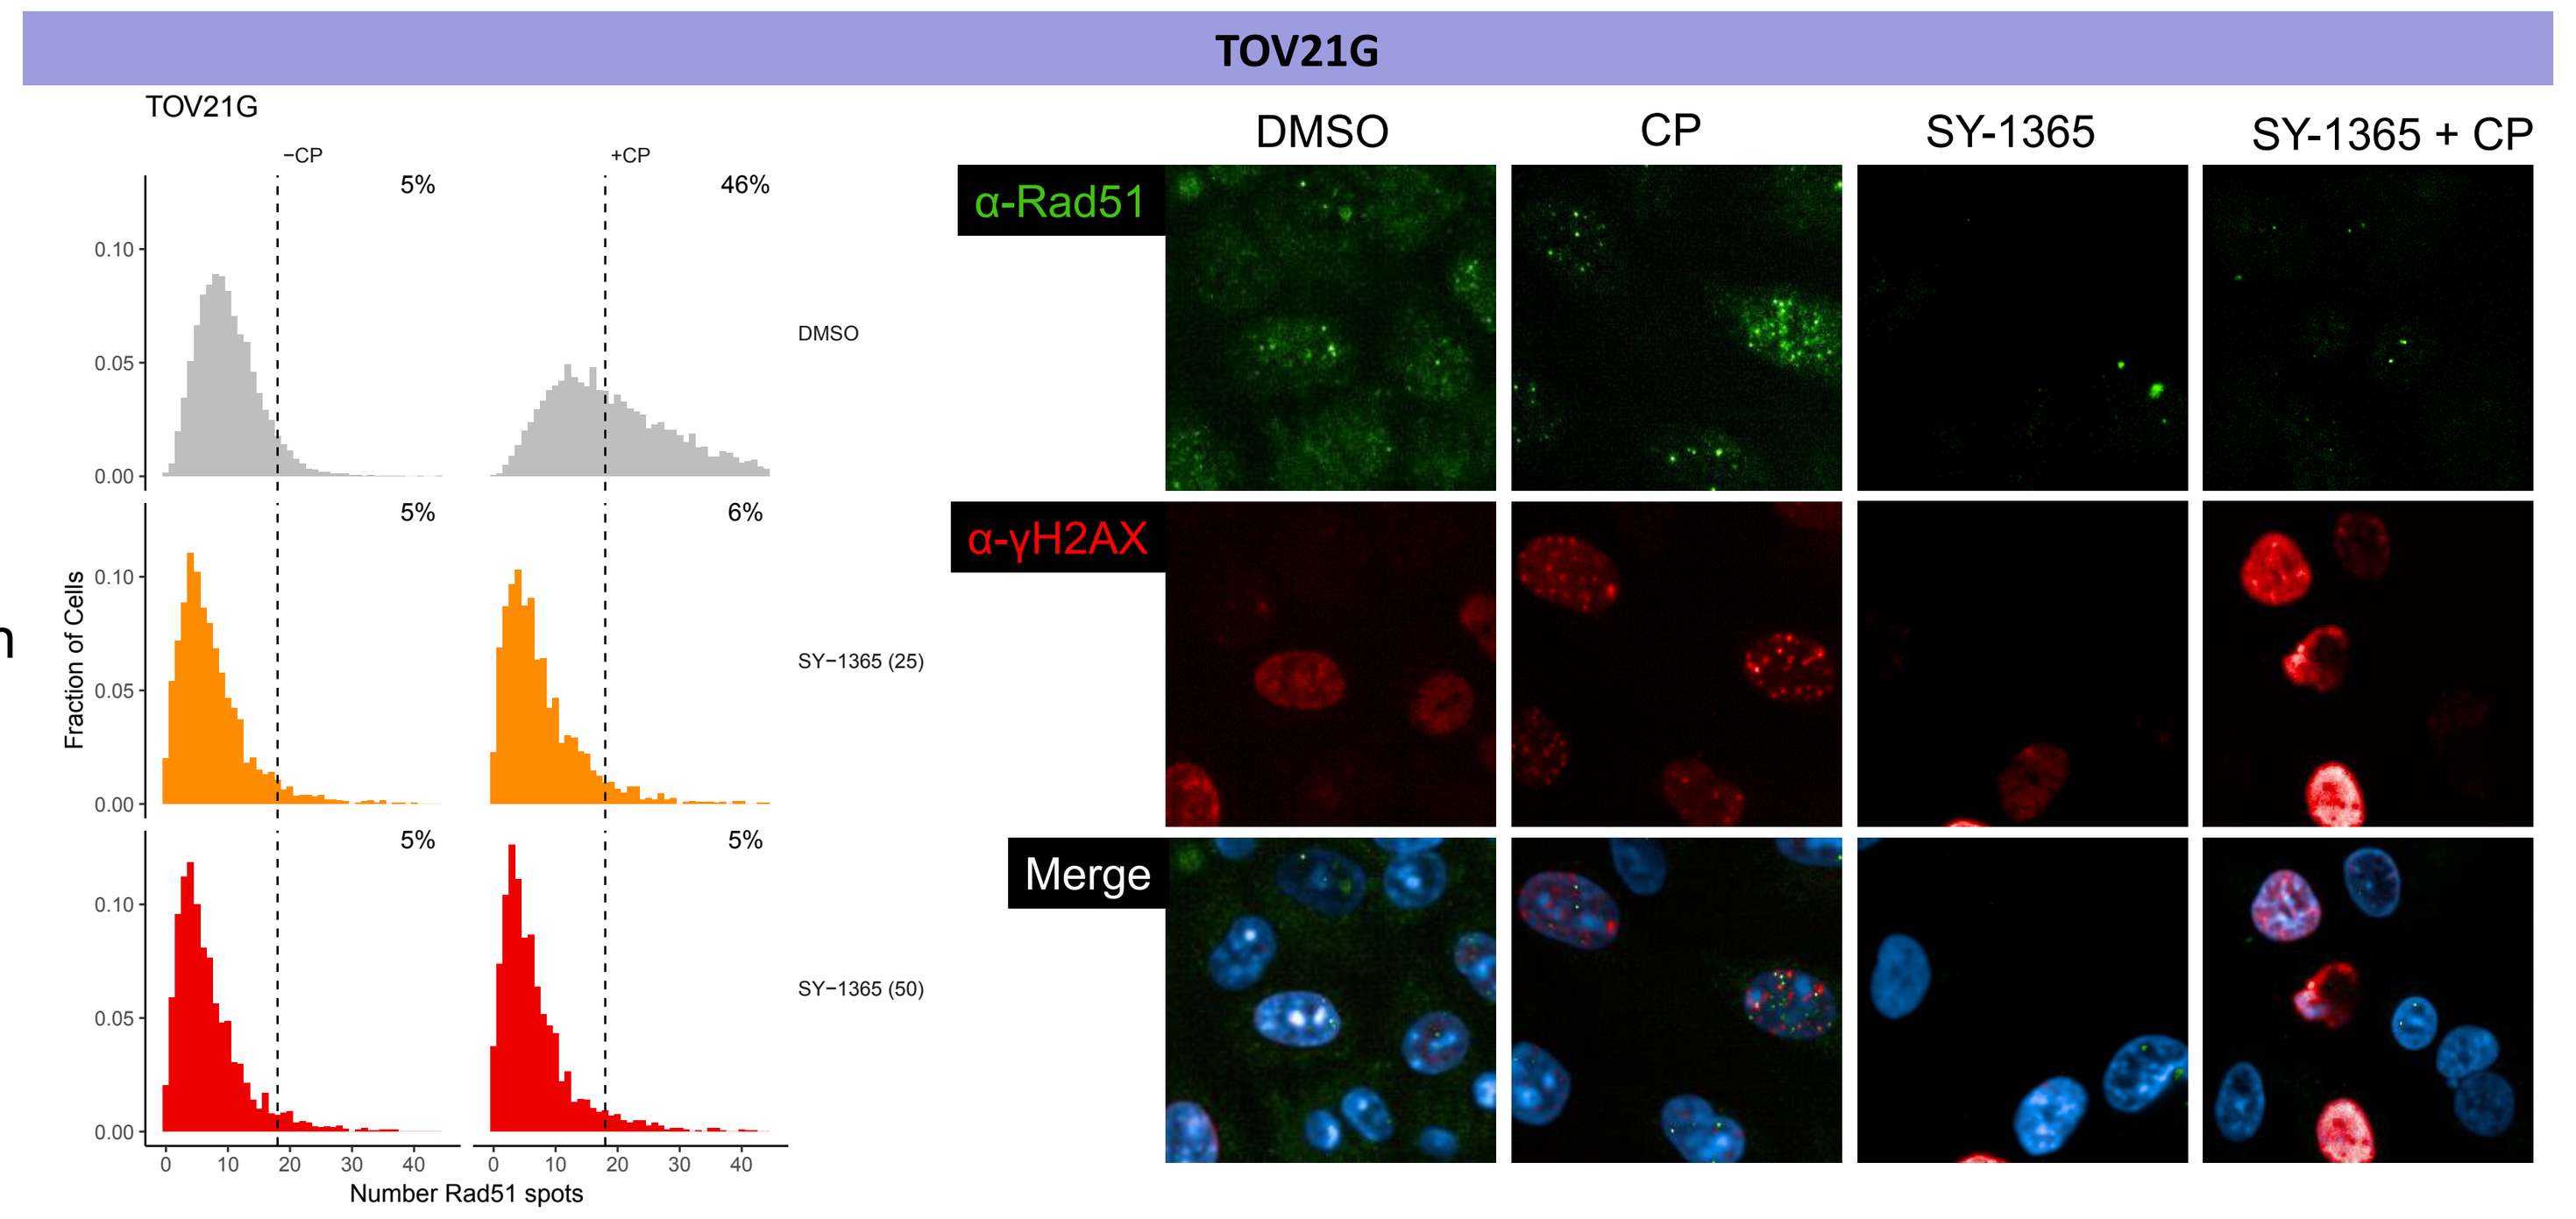
TOV21G
TOV21G
−CP
+CP
5%
46%
5%
6%
5%
5%
DMSO
SY−1365 (25)
SY−1365 (50)
Fraction of Cells
Number Rad51 spots
DMSO
CP
SY-1365
SY-1365 + CP
α-Rad51
α-γH2AX
Merge

Conclusions

- SY-1365, a selective CDK7 inhibitor, is synergistic with CP in OC preclinical models.
- SY-1365 inhibits transcription of HRR genes and inhibits DNA repair in OC preclinical models.
- These results suggest that SY-1365-mediated inhibition of HRR induces an HRD-like state resulting in enhanced sensitivity to DNA damaging agents and/or DNA repair inhibitors.
- These data support the ongoing clinical investigation of SY-1365 in combination with carboplatin in a phase 1 trial (NCT03134638).

30th EORTC-NCI-AACR Symposium, Dublin Ireland, Nov 13-16, 2018