TH-PO1152 Control of Secondary Hyperparathyroidism with Extended-release Calcifediol is Associated with Slower CKD Progression.

OPKO RENAL

Charles W. Bishop[1], Stephen A. Strugnell[1], Akhtar Ashfaq[1],
[1]OPKO Health, Miami, FL.

Introduction

Insufficient levels of serum total 25-hydroxyvitamin D (25D) increase the risk of secondary hyperparathyroidism (SHPT) in patients with stage 3 or 4 chronic kidney disease (CKD). SHPT develops and advances in most CKD patients despite aggressive treatment with cholecalciferol or ergocalciferol because serum 25D is not sufficiently raised. SHPT is associated with more rapid CKD progression and earlier dialysis. Mitigation of disease progression by effective control of SHPT has not been previously examined.

Objective

To evaluate the effect of controlling SHPT with extended-release calcifediol (ERC) on the rate of CKD progression.

Methods

Progressive changes in estimated glomerular filtration rate (eGFR) were examined post-hoc in 166 patients with vitamin D insufficiency, SHPT and stage 3-4 CKD during 1-year of treatment with ERC in pivotal trials (Sprague 2016). ERC was administered daily at 30 mcg increasing, as needed, after 12 weeks to 60 mcg to achieve a targeted ≥30% reduction in intact parathyroid hormone (iPTH). Measurements of eGFR were obtained at baseline (BL) and quarterly intervals, and 25D (DiaSorin), calcium (Ca; corrected for low albumin), phosphorus (P) and plasma iPTH (Roche Elecsys) at BL and monthly. Mean BL or quarterly iPTH levels of ≤100 pg/mL were considered "controlled" and, if maintained at 4 of these 5 assessments, "consistently controlled".

Study Population

The demographic and baseline characteristics of the study population are shown in Table 1. The population was appropriately balanced for gender and race, and the mean age, body weight, body mass index (BMI) and key biochemical parameters were representative of United States patients with stage 3-4 CKD.

Table 1. Demographics and Baseline Characteristics

Demographics	
Number of Subjects	166
Gender	
Female	84 (50.6%)
Male	82 (49.4%)
Race	
White	112 (67.5%)
African American	50 (30.1%)
Other	4 (2.4%)
Ethnicity	
Not Hispanic	133 (80.1%)
Hispanic	33 (19.9%)
Baseline Characteristics	
Weight (kg)†	100.7 (1.9)
BMI (kg/m^2)	35.5 (0.6)
Age (yrs)	65.7 (0.8)
Serum Ca (mg/dL)	9.2 (0.03)
Serum P (mg/dL)	3.8 (0.04)
Plasma iPTH (pg/mL)	146.6 (4.7)
Serum Total 25D (ng/mL)	20.1 (0.4)
eGFR (mL/min/1.73m^2)	29.6 (0.7)

†Data are Mean (SE)

Figure 1. Changes in Serum 25D, Plasma iPTH and eGFR

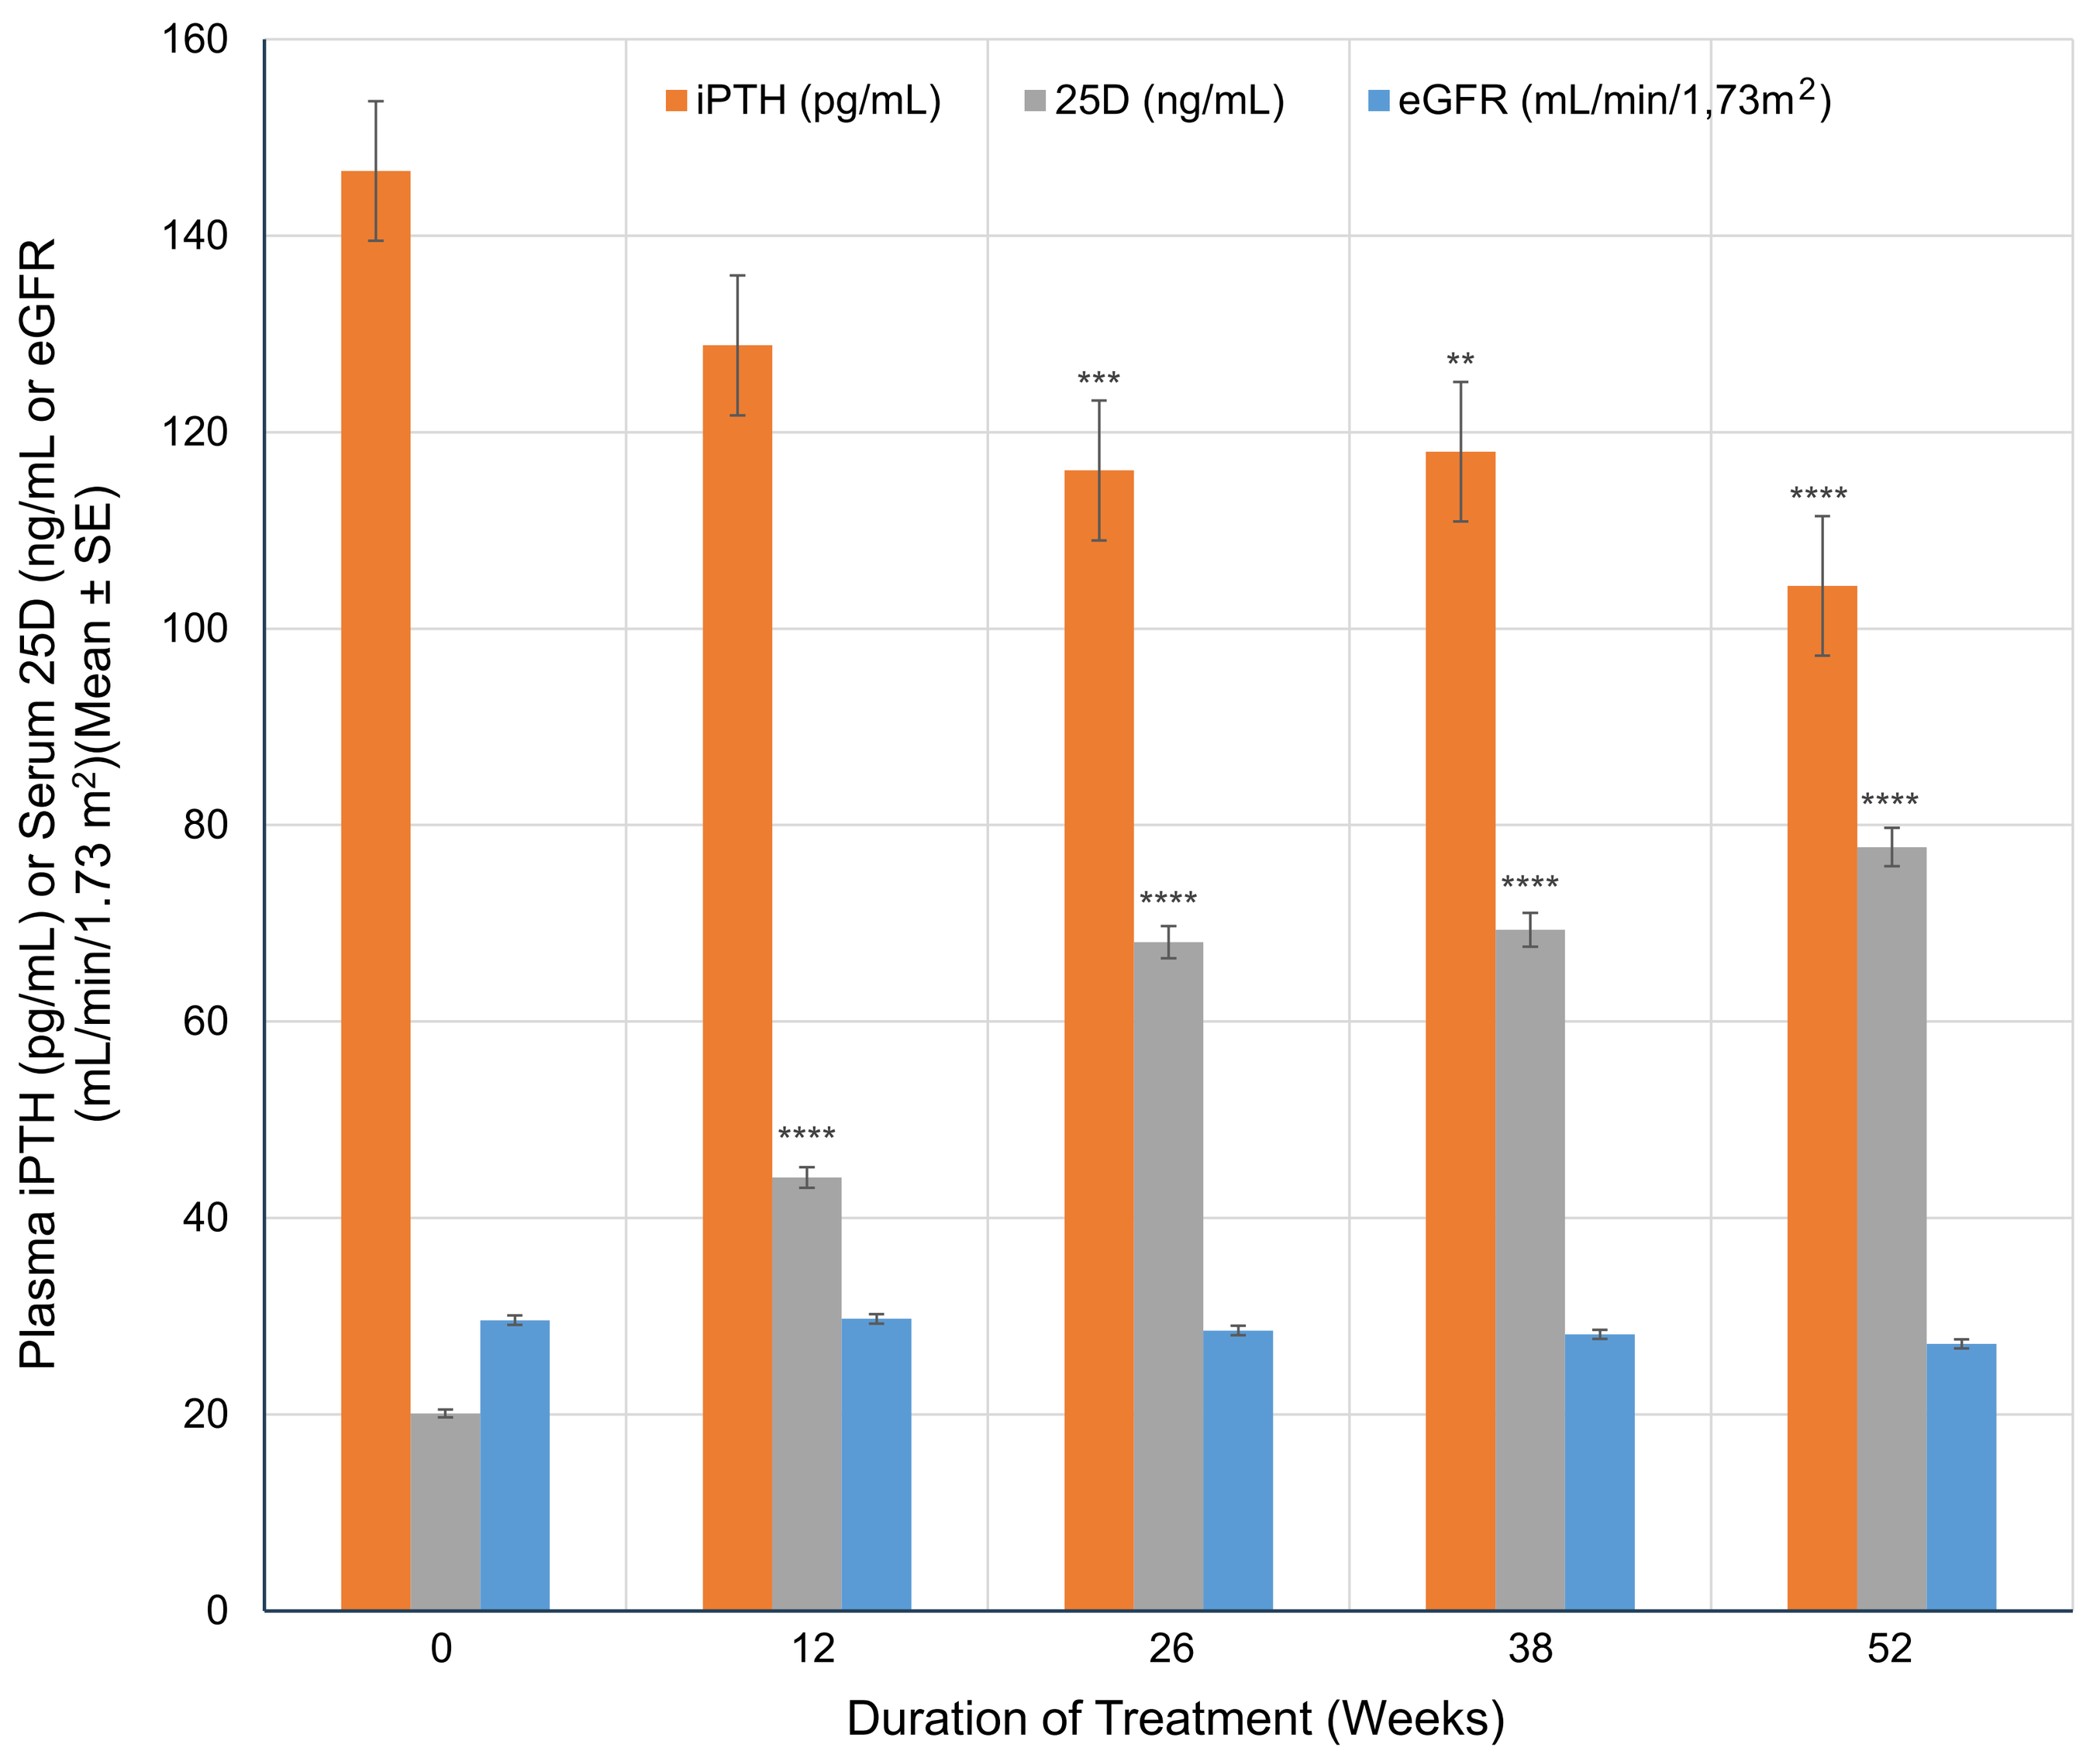
iPTH (pg/mL)
25D (ng/mL)
eGFR (mL/min/1,73m2)
Plasma iPTH (pg/mL) or Serum 25D (ng/mL or eGFR (mL/min/1.73 m2)(Mean ± SE)
Duration of Treatment (Weeks)
0
12
26
38
52

**Significantly different from baseline by ANOVA, p < 0.01
***Significantly different from baseline by ANOVA, p < 0.001
****Significantly different from baseline by ANOVA, p < 0.0001

Figure 2. Changes in Mean iPTH vs Mean 25D by eGFR Category

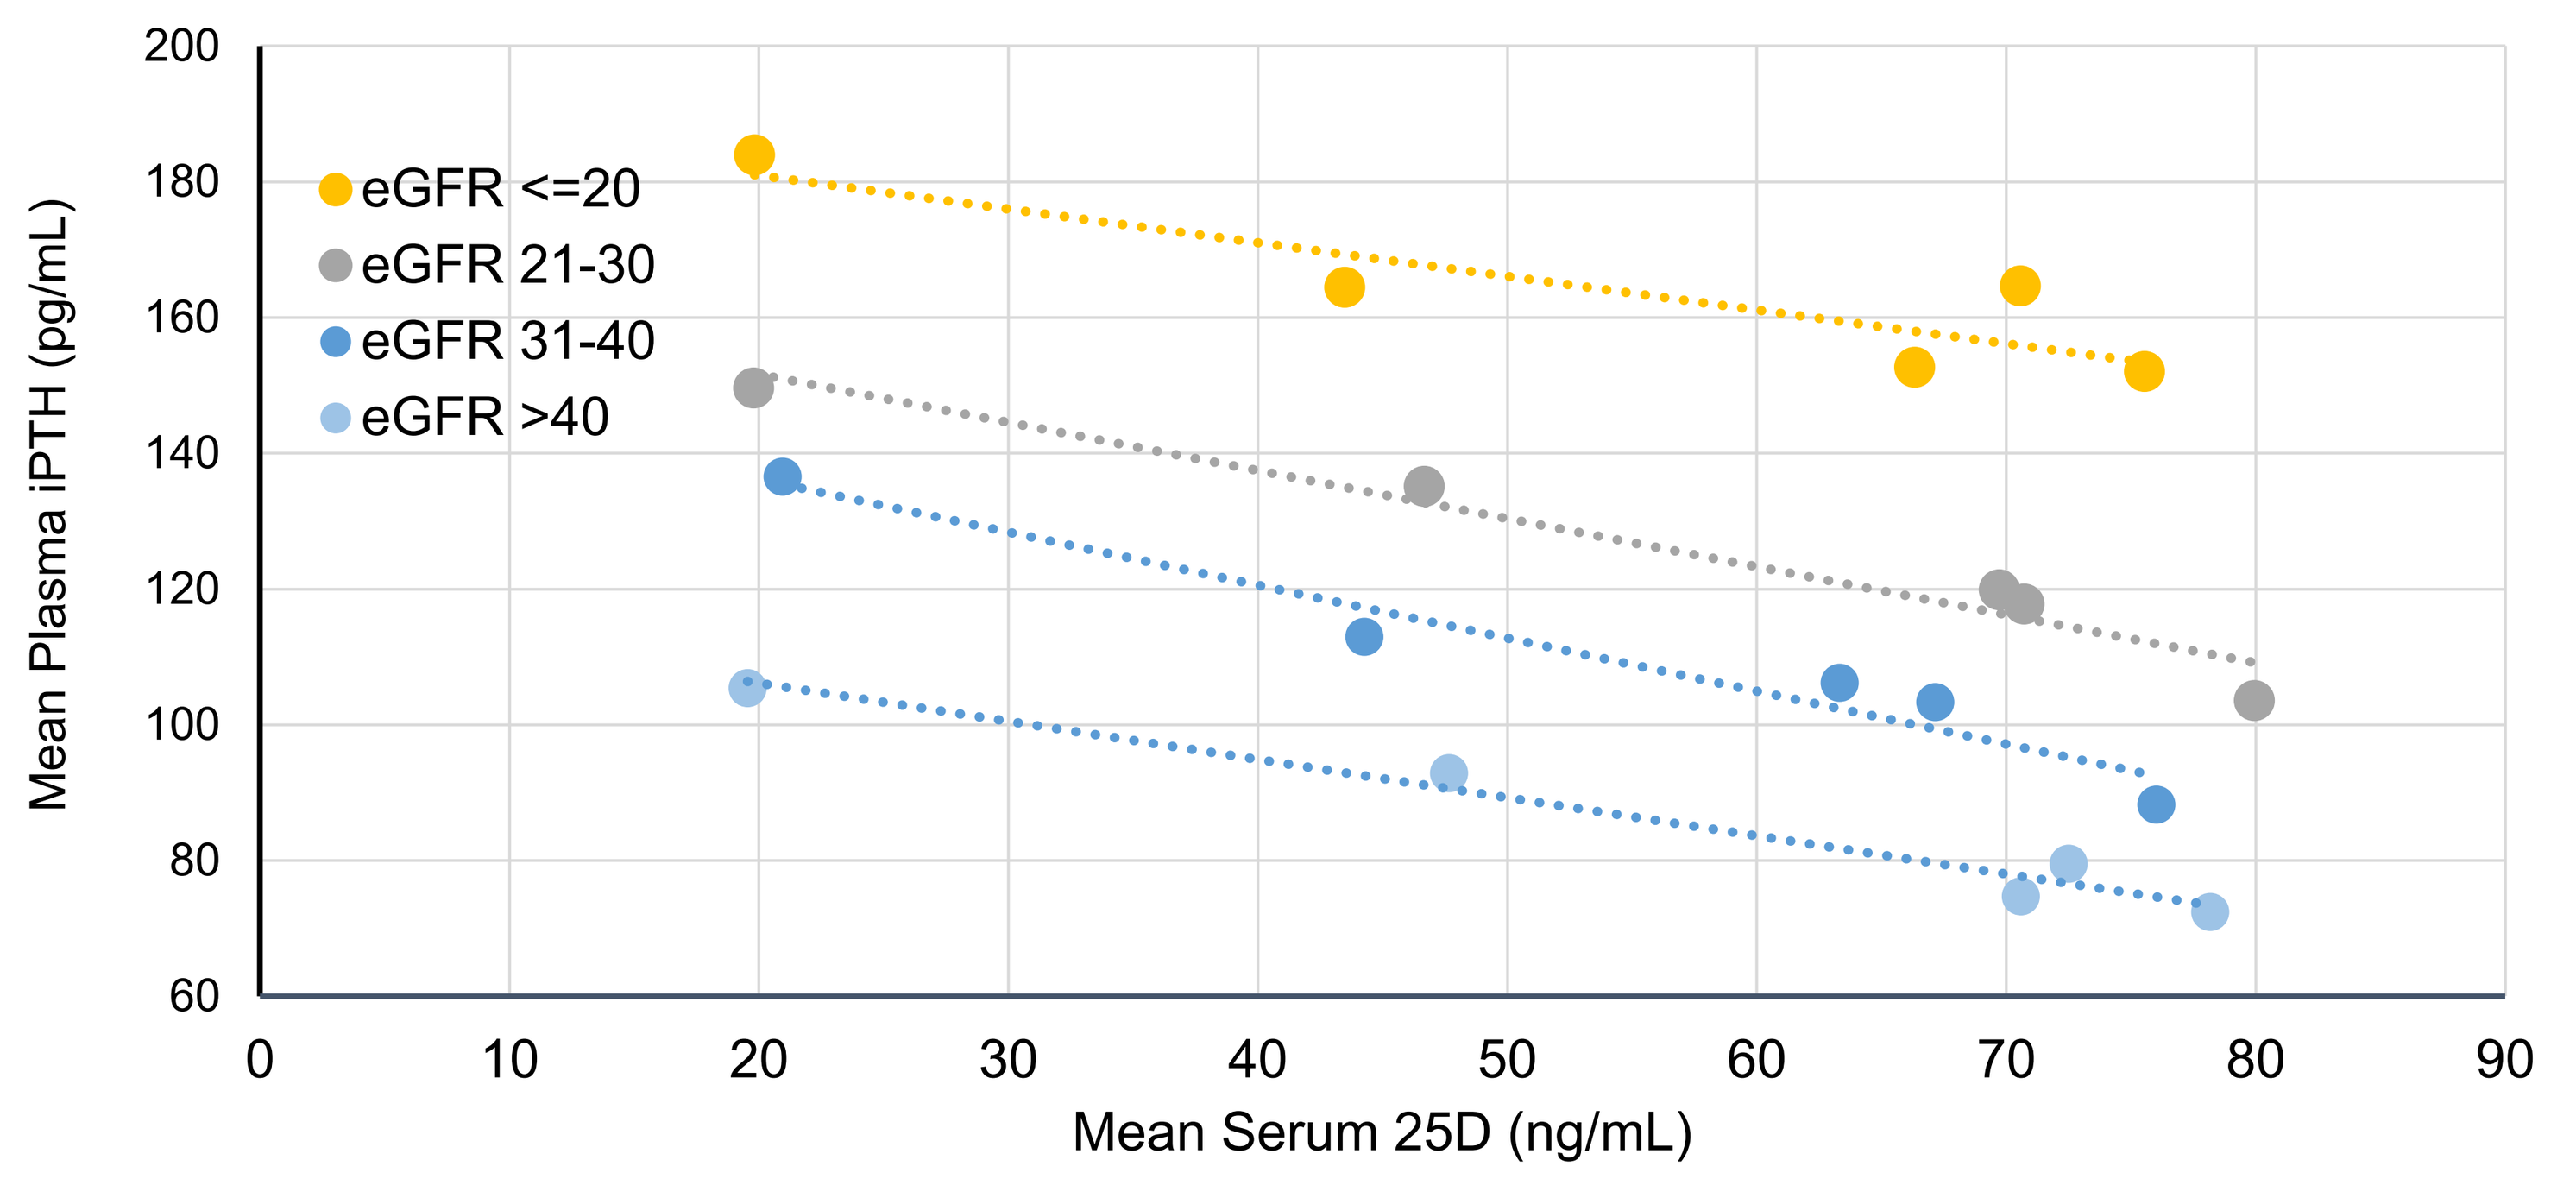
eGFR <=20
eGFR 21-30
eGFR 31-40
eGFR >40
Mean Plasma iPTH (pg/mL)
Mean Serum 25D (ng/mL)

Figure 3. Mean 52-wk eGFR Decline vs Duration of iPTH Control

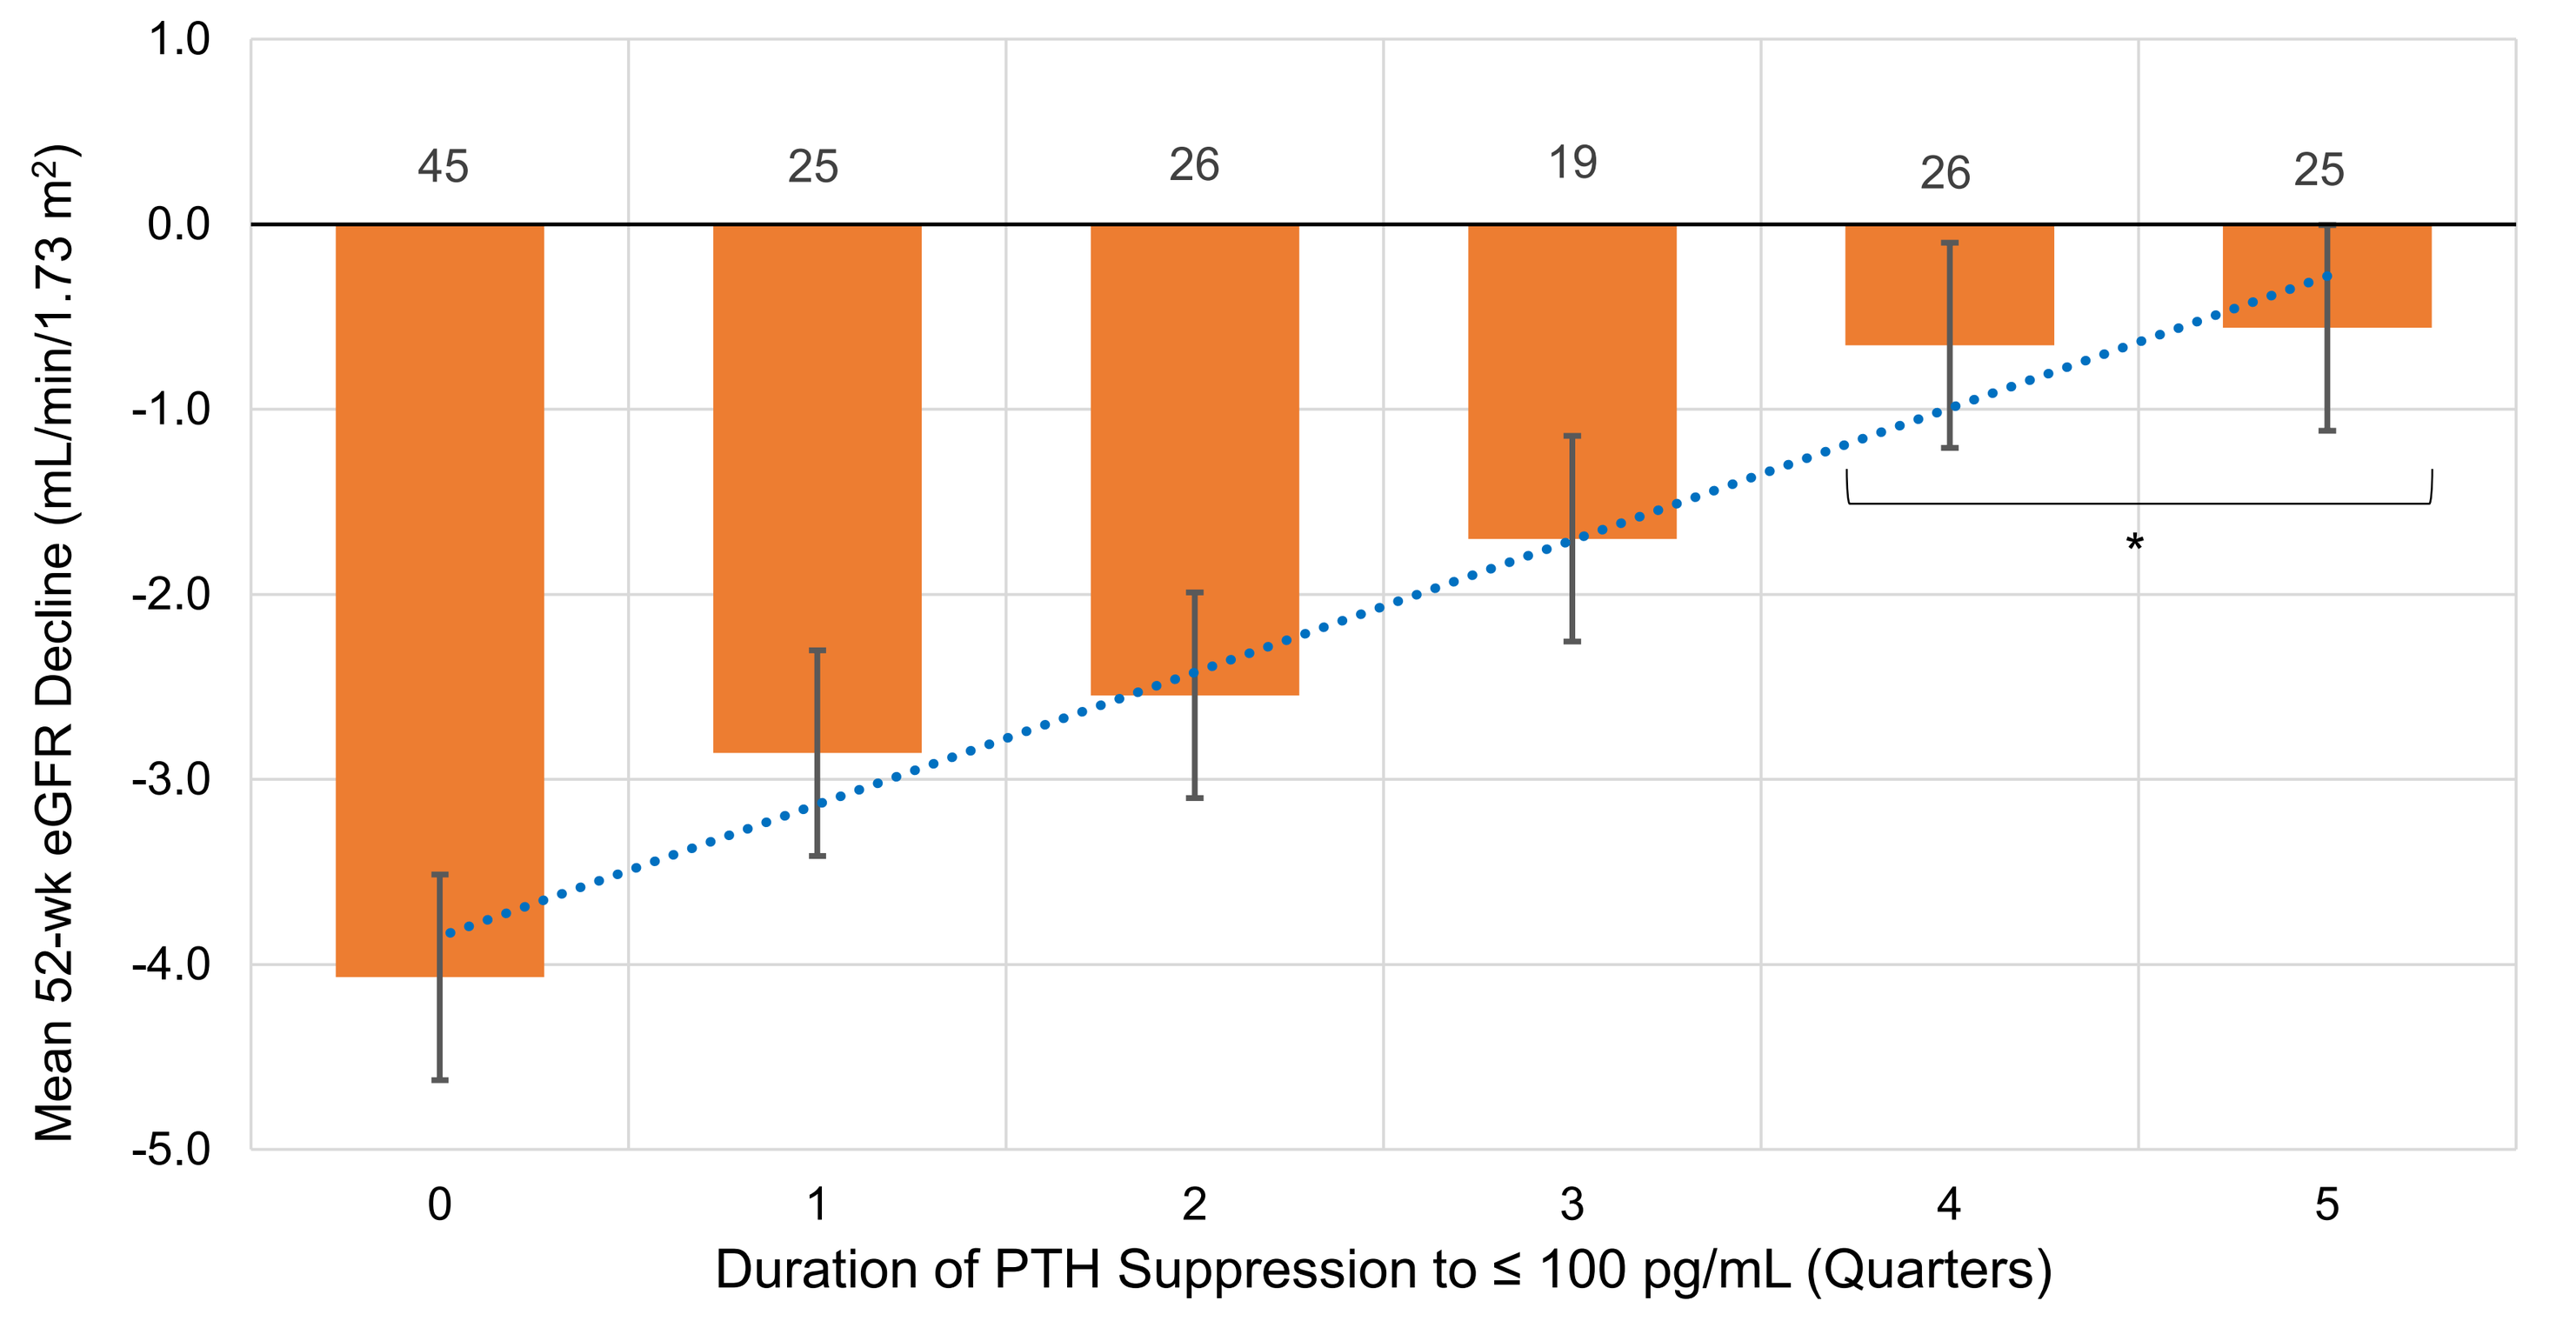
45
25
26
19
26
25
Mean 52-wk eGFR Decline (mL/min/1.73 m2)
Duration of PTH Suppression to ≤ 100 pg/mL (Quarters)

*Significantly different from 0 Quarters by t-test, p < 0.05

Figure 4. Prevalence of Increased eGFR at EOT vs Duration of iPTH Control

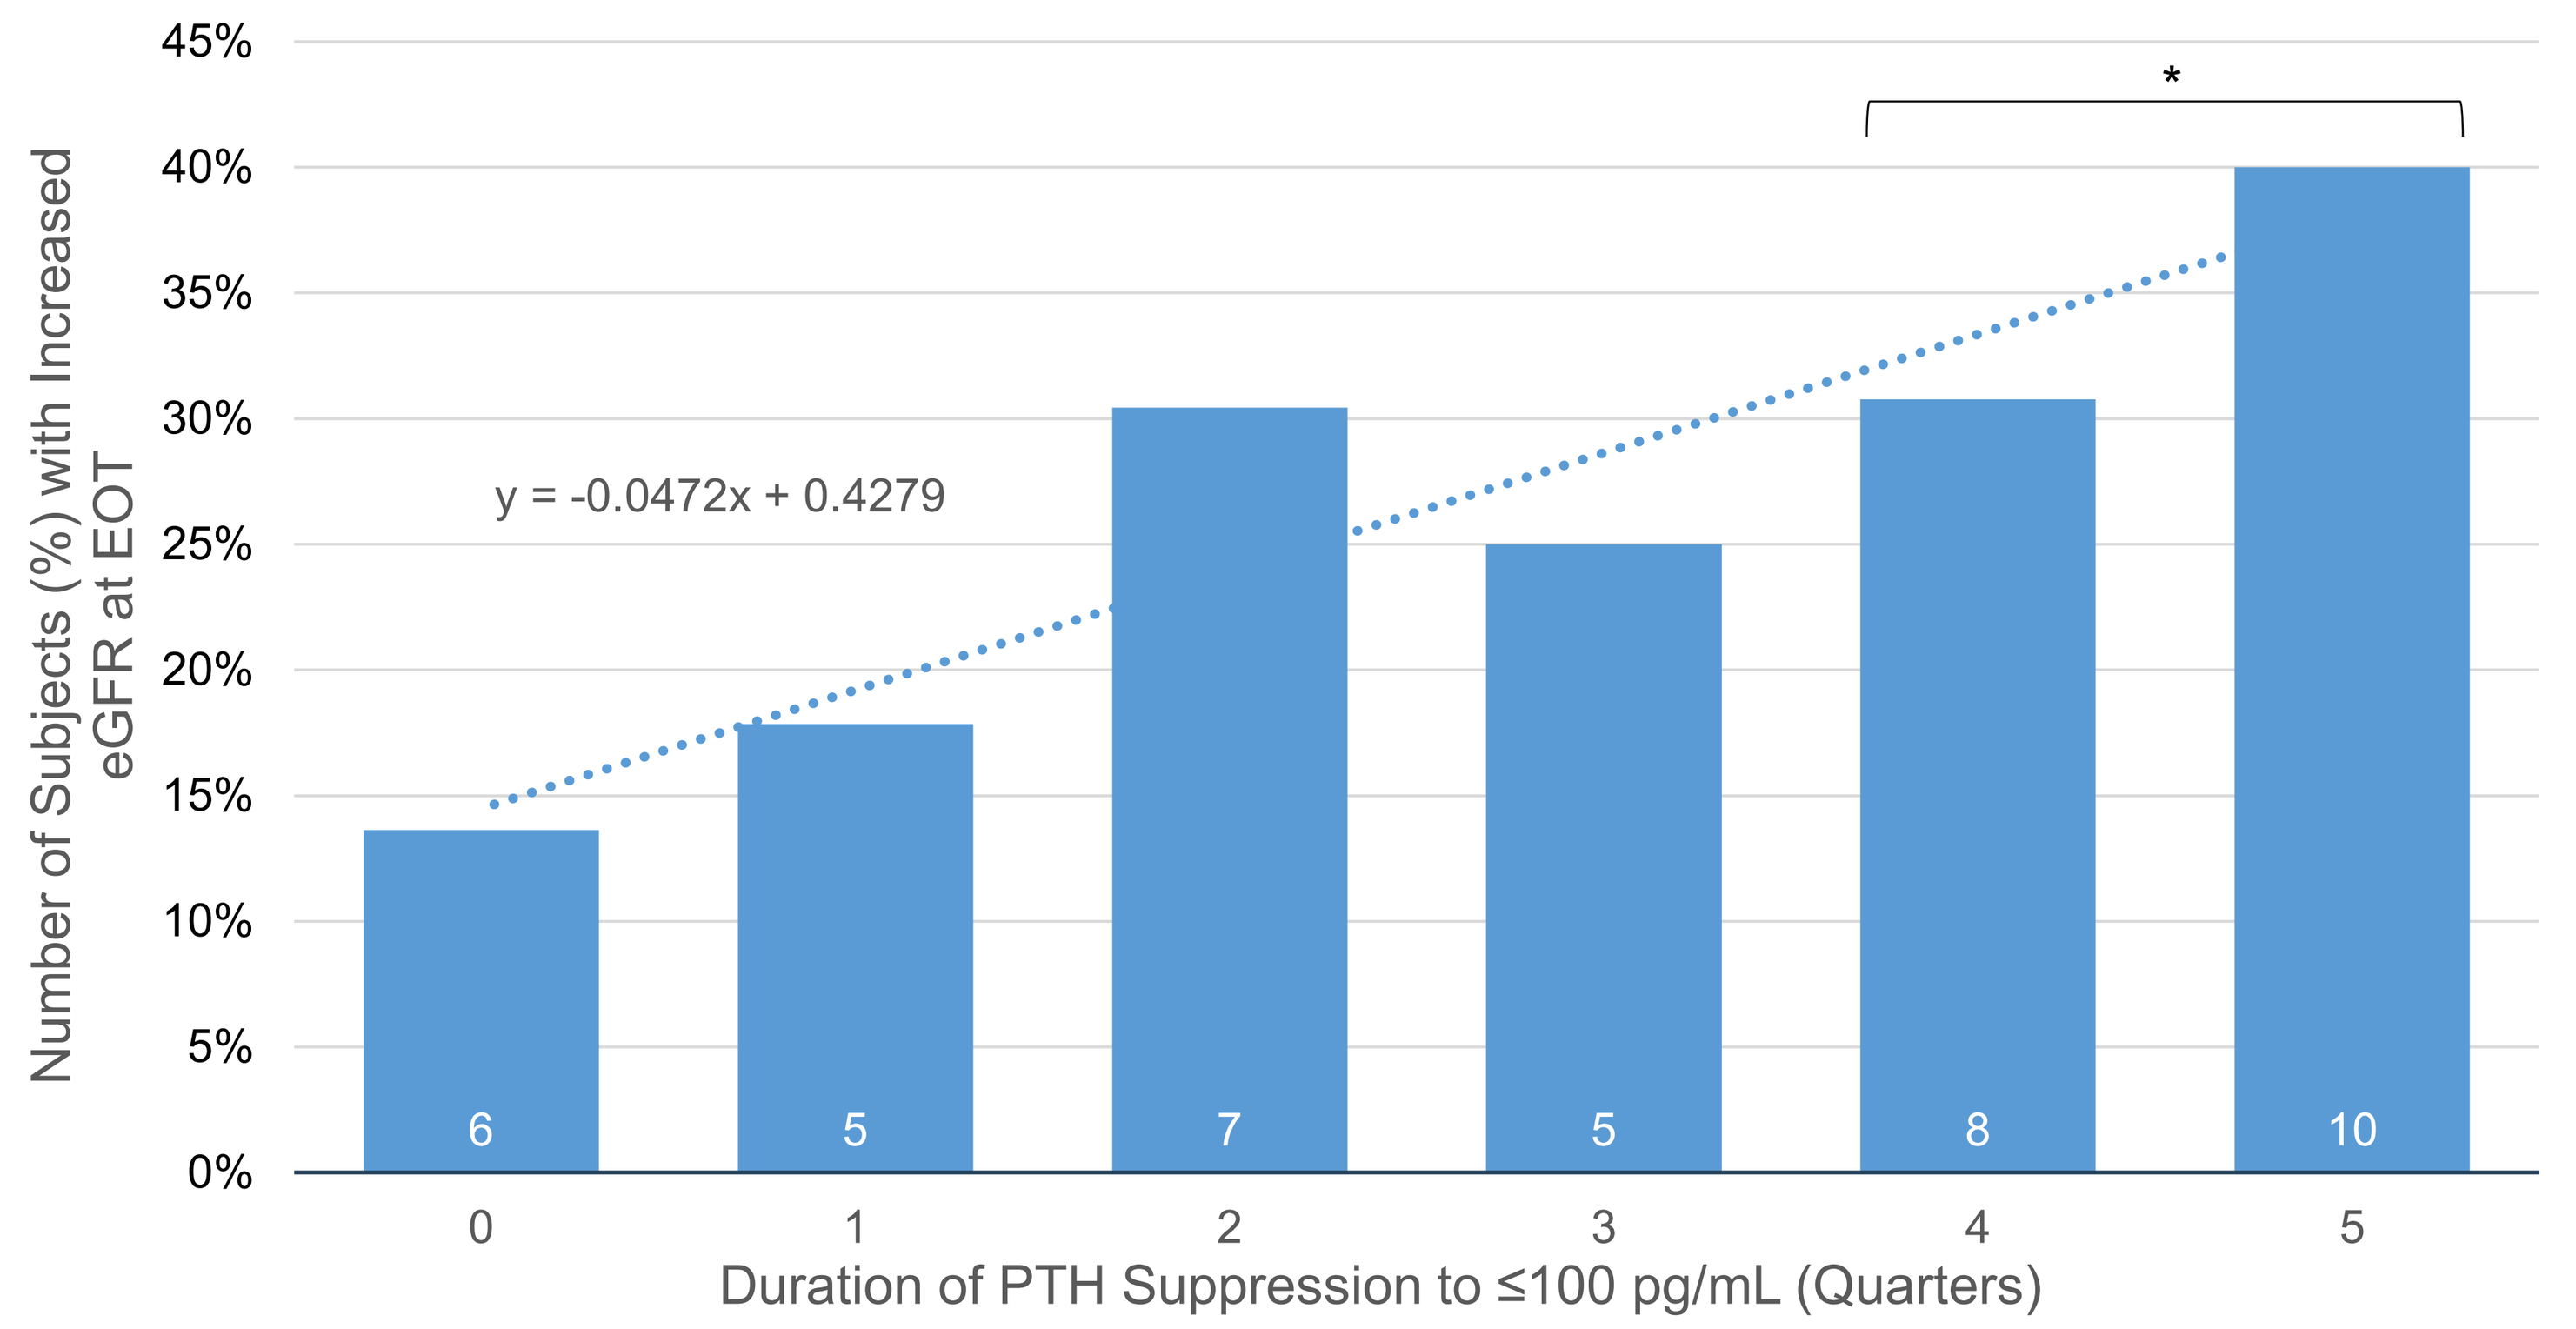
y = -0.0472x + 0.4279
Number of Subjects (%) with Increased eGFR at EOT
6
5
7
5
8
10
Duration of PTH Suppression to ≤100 pg/mL (Quarters)

*Significantly different from 0 Quarters by Chi-square test, p < 0.05

Results

- ERC treatment increased mean (±SE) serum 25D from 20.1±0.4 ng/mL at BL to 77.8±2.0 at end of treatment (EOT; p<0.0001) and decreased mean iPTH from 146.6 ± 4.7 pg/mL at BL to 104.4±6.5 (p<0.0001) without clinically meaningful changes in mean serum Ca or P (Figure 1).
- Decreases in mean iPTH were unaffected by BL eGFR (Figure 2).
- Average eGFR decline was 2.3±0.5 mL/min/1.73m^2 over the 1-year treatment period (Figure 1) but differed significantly and proportionally with duration of iPTH control (Figure 3), being greatest (4.1±0.7) in subjects who never achieved control (n=44) and least (0.61±1.2) in subjects achieving consistent control (n=51; p<0.05).
- The number of subjects having an increased eGFR by EOT rose (p < 0.05) in proportion to the duration of iPTH control achieved (Figure 4), from 6 (no control) to 18 (consistent control).

Conclusions

- A post-hoc analysis of pivotal clinical trial data with ERC indicates that early, sustained and effective treatment of SHPT is associated with mitigation of eGFR decline in patients with insufficient 25D and stage 3-4 CKD.
- Prospective studies with ERC are warranted to confirm these findings.

References

1. Sprague SM, Crawford PW, Melnick JZ et al. Use of extended-release calcifediol to treat secondary hyperparathyroidism in stage 3 and 4 chronic kidney disease. Am J Nephrol 2016;44:316-325.

Acknowledgements

This study was sponsored by the Renal Division of OPKO Health Inc.

FOR MORE INFORMATION PLEASE CONTACT:
Stephen A. Strugnell, PhD, OPKO Health, Inc. 4400 Biscayne Blvd. Miami, FL 33137.
Email: sstrugnell@opko.com