Presentation #48
ASM-ESCMID 9/5/2019

Time-kill Kinetics of the Novel Antibacterial Agent ACX-362E against *Clostridioides difficile*

B Murray[1], C. Wolfe[1], A Marra[1], M. H. Silverman[2], C Pillar[1], D Shinabarger[1]

[1]Micromyx, Kalamazoo, MI; [2]Acurx, LLC, White Plains, NY

- Micromyx., Inc.
- amarra@Micromyx.com

ABSTRACT

Background: ACX-362E is a novel DNA polymerase IIIC inhibitor in clinical development for the treatment of *Clostridioides difficile* infections. The bactericidal activity of ACX-362E was evaluated against actively-growing *C. difficile* by determining the MIC and the minimum bactericidal concentration (MBC) against three *C. difficile* isolates. Time-kill kinetics assays were additionally performed against the three *C. difficile* isolates with metronidazole and vancomycin as comparators.

Methods: ACX-362E and comparator susceptibility testing was conducted per CLSI published methodology (M11). MBC values were obtained by determining the viable count of each inoculum immediately following inoculation of MIC plates and comparing that to the viable count in the MIC well and three wells above the MIC post-incubation. The MBC was then defined as the lowest concentration of agent to demonstrate a 99% killing of bacteria (CLSI M26-A). Time-kill kinetics were determined using methods described by CLSI (M26-A) used to evaluate the bactericidal activity of test agents. All compounds were tested in the assay at 8X, 16X and 32X the broth microdilution MIC and viable counts were determined at 2, 6, 24 and 48 hours.

Results: ACX-362E had MIC values of 1 µg/mL against *C. difficile* MMX 5680 (ribotype 027), BAA 1382 (ribotype 078), and BAA 1875 (ribotype 078), respectively, and corresponding MBC:MIC ratios of 1, 2 and >8. MBC:MIC ratios for metronidazole were 1, 2 and 2 and for vancomycin against the same three *C. difficile* isolates they were 1, 2 and 1, respectively. When the time-kill kinetics of ACX-362E were evaluated against two isolates of *C. difficile*, bactericidal activity was observed at 24 and 48 hr at 16X and 32X the MIC. Against the third isolate, ACX-362E did not demonstrate the ≥3 $\log_{10}$ CFU/mL killing required for bactericidal activity but bacterial levels were reduced by >2 $\log_{10}$ CFU/mL at the later time points at the 16X and 32X the MIC concentrations.

Conclusions: Against two of three isolates, the bactericidal activity of ACX-362E was observed to occur at concentrations similar to its inhibitory activity, as demonstrated by MBC:MIC ratios and reflected in time-kill kinetics assays. This ability bodes well for the therapeutic potential of ACX-362E, the first DNA polymerase IIIC inhibitor to enter human trials. to suppress and kill *C. difficile*.

INTRODUCTION

- The Gram-positive anaerobic spore-forming bacterium *Clostridioides difficile* is the most frequently-reported nosocomial pathogen, recently surpassing *Staphylococcus aureus* for infections in the US (1).
- A 2011 surveillance study reported that 453,000 cases and 29,000 deaths were associated with *C. difficile*; approximately 25% of these were community-acquired (2).
- Relapse/recurrence post-treatment with therapies such as metronidazole (MET) and vancomycin (VAN) are frequent (3).
- The severity and prevalence of these infections have caused the CDC to identify *C. difficile* as an "urgent threat" (4)
- ACX-362E is being developed for oral treatment of *C. difficile* infections (CDI).
- ACX-362E is a dichlorobenzyl purine analog with a unique mechanism of action that binds and inhibits DNA polymerase IIIC from aerobic, low G+C Gram-positive bacteria. ACX-362E has been shown to directly inhibit the DNA pol IIIC from *C. difficile*, with a K_i of 0.325 µM (5).
- DNA pol III is absent in Gram-negative bacteria and thus, ACX-362E lacks activity against most of the organisms that make up the colonic flora, enhancing its profile as a selective treatment for CDI.

OBJECTIVE

- In this study, the bactericidal activity of ACX-362 was evaluated against *C. difficile* isolates in comparison with vancomycin (VAN) and metronidazole (MET).

METHODS

- Test organisms were obtained from the American Type Culture Collection (ATCC, Manassas, VA) and had been isolated from infected patients and determined to be toxin producers of different ribotypes.
- MIC values were determined by broth microdilution following guidelines .recommended by the Clinical and Laboratory Standards Institute (CLSI; M07-A7, M11-A9 and M100-A29). MBC determinations were performed as recommended in CLSI document M26.
- Time-kill kinetics assays were conducted per CLSI M26, and all compounds were evaluated at 8X, 16X and 32X the broth microdilution MIC. Cultures in the time-kill assay were sampled at 0, 2, 6, 24, and 48 hours.

RESULTS

- The time-kill kinetics for ACX-362E, VAN and MET against *C. difficile* MMX 5680 are shown in **Figure 1**, the corresponding change in $\log_{10}$ CFU/mL relative to baseline are shown in **Table 2**. For ACX-362E, bactericidal activity was observed at 16X and 32X the MIC at 24 and 48 hours.
- ACX-362E had an MBC:MIC ratio of 1 against *C. difficile* MMX 5680 (ribotype 027 isolate), as did MET and VAN.
- **Figure 2** shows the time-kill kinetics for ACX-362E, VAN and MET against *C. difficile* BAA-1382, and **Table 3** displays the change in $\log_{10}$ CFU/mL relative to baseline. All three tested concentrations of ACX-362E demonstrated bactericidal activity at 24 and 48 hours.
- ACX-362E had an MBC:MIC ratio of 2 against *C. difficile* BAA 1382 (ribotype 078 isolate), as did MET and VAN.
- Time-kill kinetics for ACX-362E, VAN and MET are shown in **Figure 3** and **Table 4** displays the change in $\log_{10}$ CFU/mL relative to baseline. ACX-362E did not demonstrate bactericidal activity as defined by a 3-log kill but by the 24 and 48 hr time points exhibited >2-log killing relative to the starting inoculum.
- ACX-362E had an MBC:MIC ratio of >8 against *C. difficile* BAA 1875 (ribotype 078 isolate); ratios for MET and VAN were 2 and 1, respectively; these drugs were also bactericidal by time-kill.

RESULTS

- The MIC and MBC values for ACX-362E, metronidazole and vancomycin are shown for triplicate independent inocula of the three *C. difficile* isolates in **Table 1.**
- ACX-362E demonstrated an MIC of 1 µg/mL for all three isolates and for each replicate.
- For MET and VAN, the highest MIC value obtained for the triplicate testing was used to calculate 8X, 16X, and 32X the MIC for the time-kill study.

Table 2. Time-kill Kinetic Data for ACX-362E, Vancomycin (VAN) and Metronidazole (MET) against *C. difficile* MMX 5680

Drug/test concentration	ΔLog CFU/mL relative to 0 hr			
	2	6	24	48
ACX-362E 8X MIC	-1.02	-1.25	-0.90	0.17
ACX-362E 16X MIC	-0.93	-1.50	-3.11	-4.96
ACX-362E 32X MIC	-1.22	-1.86	-3.27	-4.96
VAN 8X MIC	-1.18	-1.30	-3.30	-4.96
VAN 16X MIC	-1.04	-1.47	-3.30	-4.48
VAN 32X MIC	-1.33	-1.62	-3.40	-4.96
MET 8X MIC	-1.45	-4.96	-4.96	-4.96
MET 16X MIC	-1.53	-4.96	-4.96	-4.96
MET 32X MIC	-1.38	-4.96	-4.96	-4.96
no drug - water	-0.77	0.63	0.77	0.55
no drug - DMSO	-1.01	0.09	0.95	0.25

Table 3. Time-kill Kinetic Data for ACX-362E, Vancomycin (VAN) and Metronidazole (MET) against *C. difficile* BAA-1382

Drug/test concentration	ΔLog CFU/mL relative to 0 hr			
	2	6	24	48
ACX-362E 8X MIC	-0.42	-1.47	-3.39	-3.24
ACX-362E 16X MIC	-0.02	-0.83	-3.19	-3.83
ACX-362E 32X MIC	0.07	-0.69	-3.39	-4.37
VAN 8X MIC	-0.55	-0.86	-3.44	-4.67
VAN 16X MIC	-0.40	-0.63	-2.54	-3.89
VAN 32X MIC	-0.22	-0.56	-2.86	-4.07
MET 8X MIC	-4.67	-4.67	-4.67	-4.67
MET 16X MIC	-4.67	-4.67	-4.67	-4.67
MET 32X MIC	-3.67	-4.67	-4.67	-4.67
no drug - water	0.11	1.74	1.46	1.16
no drug - DMSO	0.07	1.78	0.34	0.38

Table 4. Time-kill Kinetic Data for ACX-362E, Vancomycin (VAN) and Metronidazole (MET) against *C. difficile* BAA-1875

Drug/test concentration	ΔLog CFU/mL relative to 0 hr			
	2	6	24	48
ACX-362E 8X MIC	0.10	-1.42	-1.99	1.41
ACX-362E 16X MIC	-0.02	-1.95	-2.29	-2.38
ACX-362E 32X MIC	0.06	-1.54	-2.37	-2.44
VAN 8X MIC	-0.13	-0.92	-3.20	-3.45
VAN 16X MIC	-0.21	-0.48	-2.69	-4.05
VAN 32X MIC	-0.21	-0.38	-2.34	-3.15
MET 8X MIC	-4.05	-4.05	-4.05	-4.05
MET 16X MIC	-4.05	-4.05	-4.05	-4.05
MET 32X MIC	-4.05	-4.05	-4.05	-4.05
no drug - water	0.37	1.76	0.23	0.62
no drug - DMSO	0.90	1.38	1.02	0.86

Table 1. Activity (µg/mL) of ACX-362E, Metronidazole and Vancomycin against *C. difficile* Isolates

Isolate No.	Type	Replicate	ACX-362E		MET		VAN	
			MIC	MBC	MIC	MBC	MIC	MBC
MMX 5680	Ribotype 027	A	1	1	2	2	0.5	0.5
		B	1	1	4	4	0.5	0.5
		C	1	2	2	2	0.25	0.25
BAA-1382	Ribotype 012	A	1	4	0.5	0.5	1	2
		B	1	2	0.5	1	1	1
		C	1	2	1	1	1	2
BAA-1875	Ribotype 078	A	1	>8*	0.5	1	0.25	0.5
		B	1	2	1	1	0.5	0.5
		C	1	>8*	0.5	0.5	0.5	0.5

Figure 1. Time-kill kinetic results for ACX-362E, Vancomycin (VAN) and Metronidazole (MET) against *C. difficile* MMX 5680

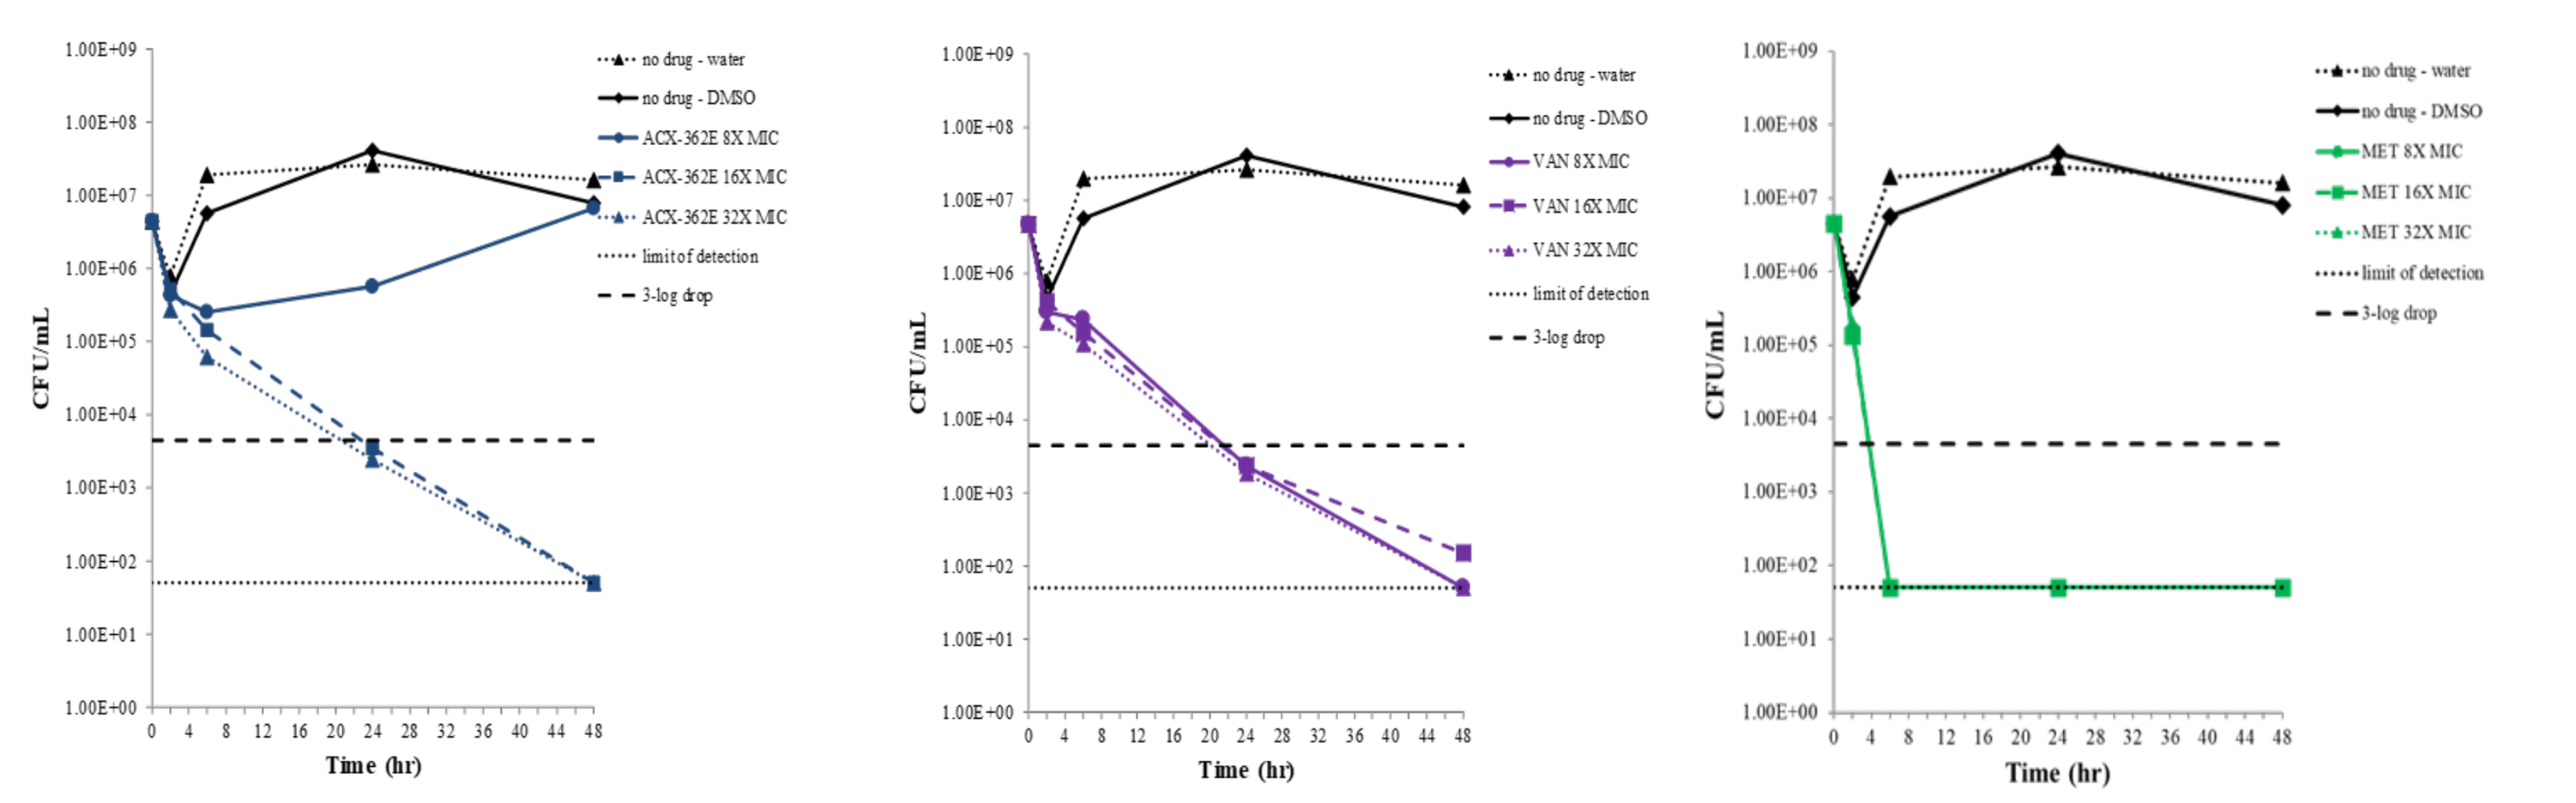

CFU/mL
Time (hr)
no drug - water
no drug - DMSO
ACX-362E 8X MIC
ACX-362E 16X MIC
ACX-362E 32X MIC
VAN 8X MIC
VAN 16X MIC
VAN 32X MIC
MET 8X MIC
MET 16X MIC
MET 32X MIC
limit of detection
3-log drop

Figure 2. Time-kill kinetic results for ACX-362E, Vancomycin (VAN) and Metronidazole (MET) against *C. difficile* BAA-1382

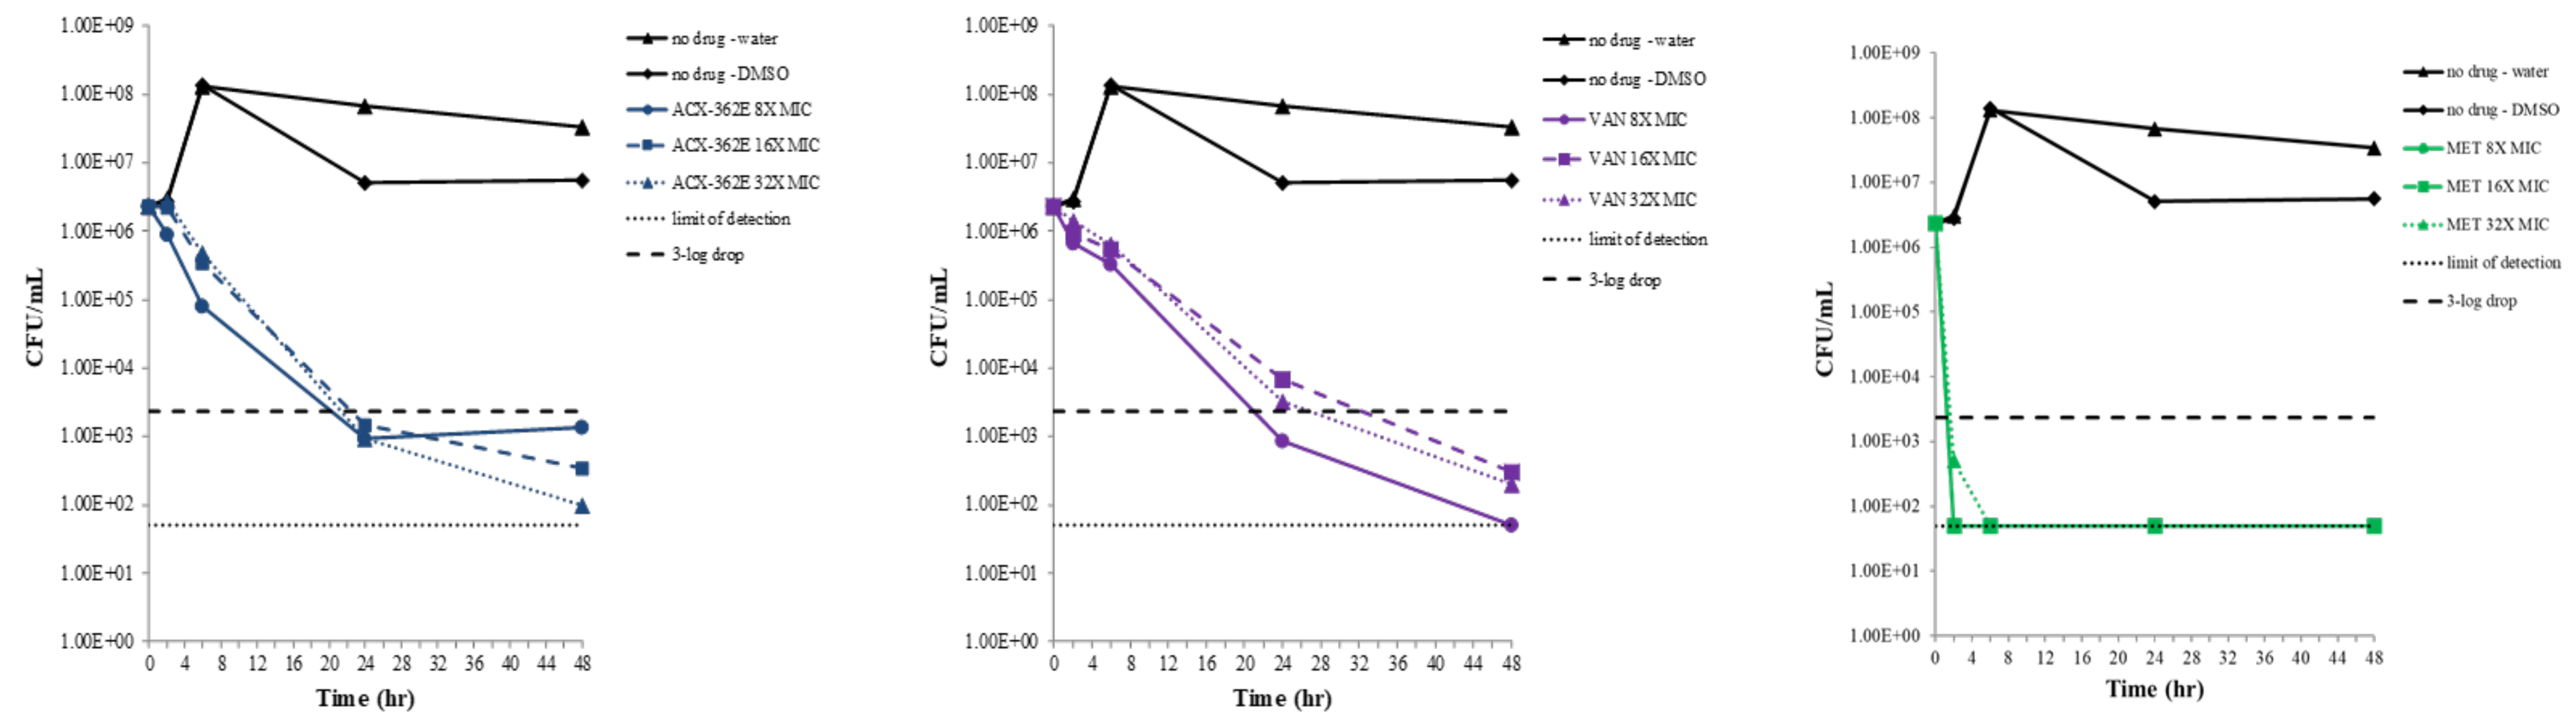

CFU/mL
Time (hr)
no drug - water
no drug - DMSO
ACX-362E 8X MIC
ACX-362E 16X MIC
ACX-362E 32X MIC
VAN 8X MIC
VAN 16X MIC
VAN 32X MIC
MET 8X MIC
MET 16X MIC
MET 32X MIC
limit of detection
3-log drop

Figure 3. Time-kill kinetic results for ACX-362E, Vancomycin (VAN) and Metronidazole (MET) against *C. difficile* BAA-1875

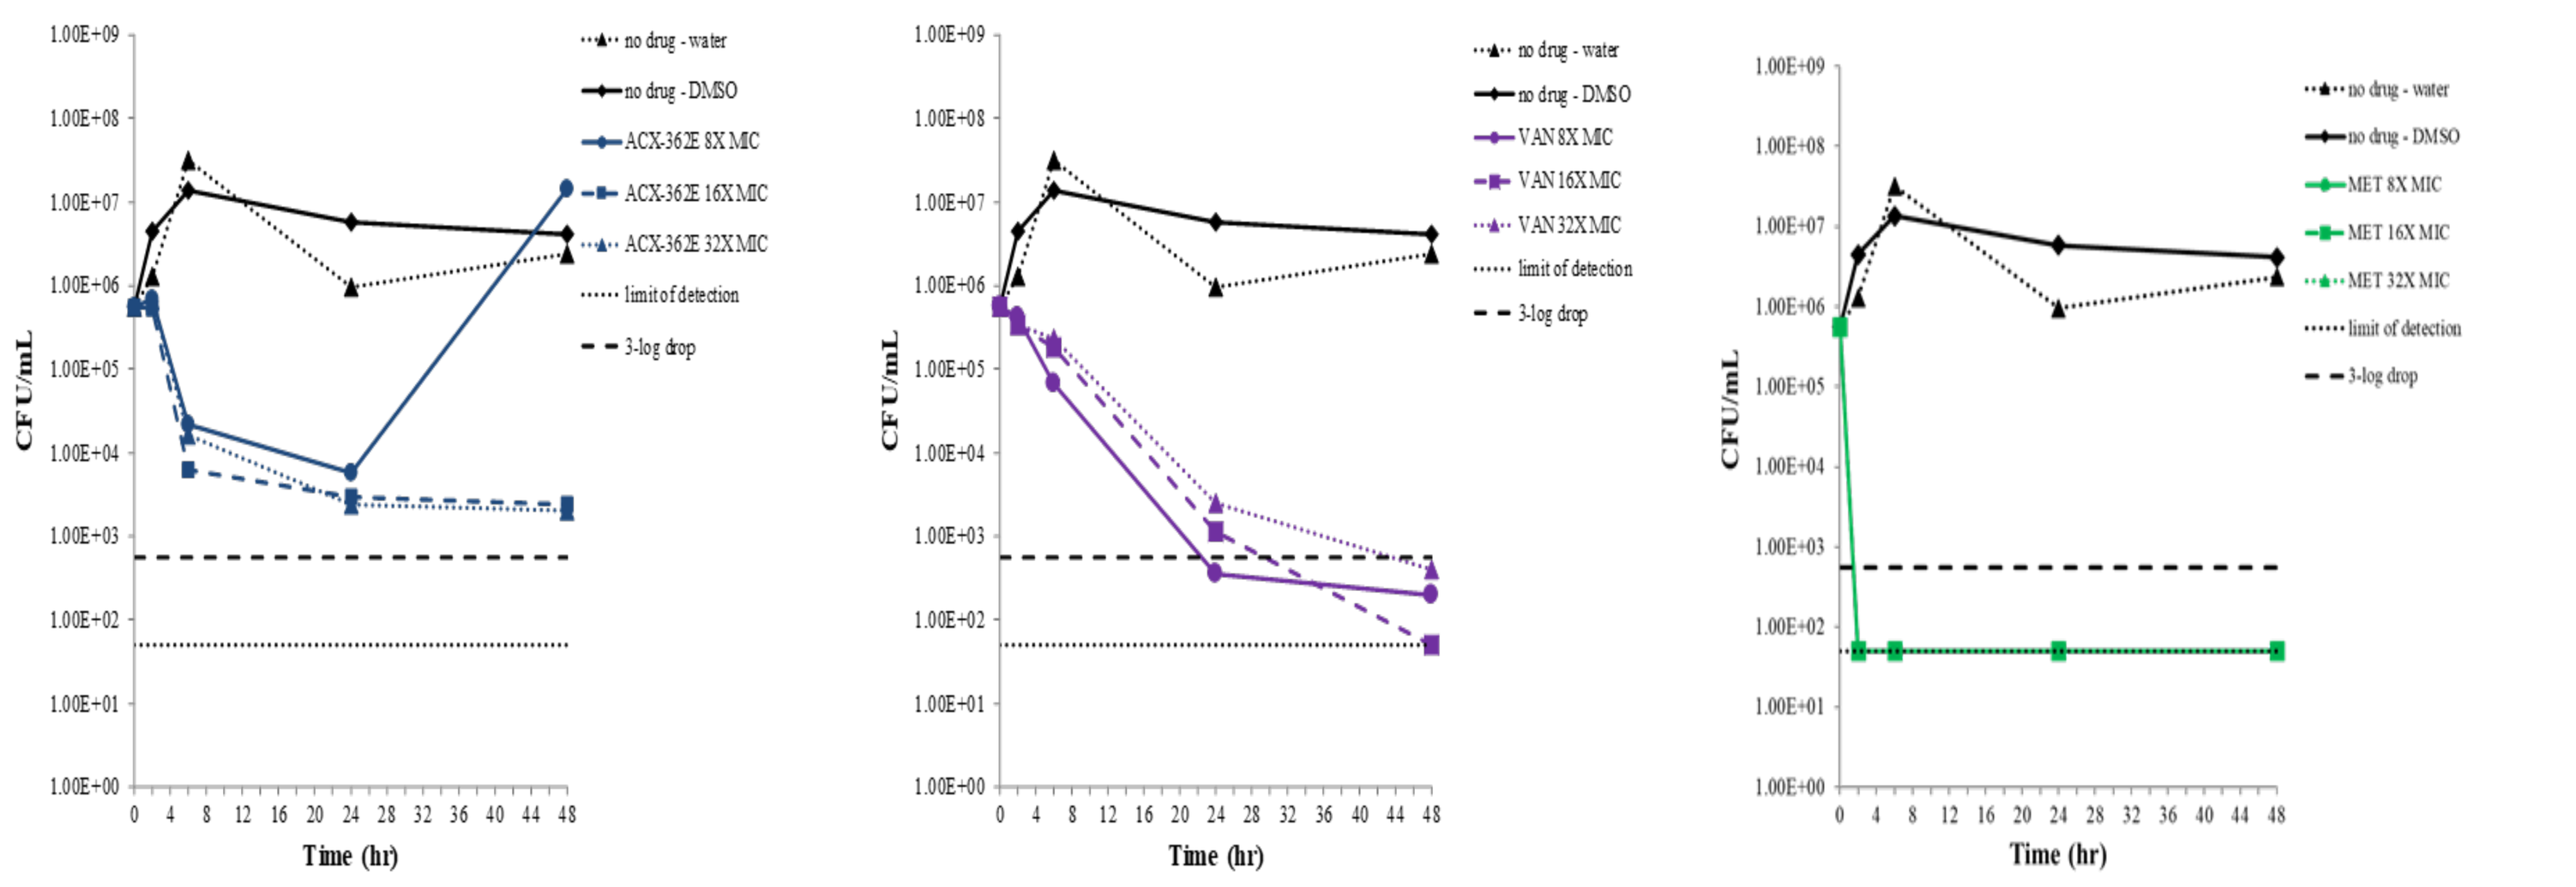

CFU/mL
Time (hr)
no drug - water
no drug - DMSO
ACX-362E 8X MIC
ACX-362E 16X MIC
ACX-362E 32X MIC
VAN 8X MIC
VAN 16X MIC
VAN 32X MIC
MET 8X MIC
MET 16X MIC
MET 32X MIC
limit of detection
3-log drop

CONCLUSIONS

- Against two of the three *C. difficile* isolates, ACX-362E had MBC:MIC ratios of 1 to 4 across replicates indicating bactericidal activity.
- For the remaining isolate, MBC:MIC ratios of 2 and >8 were observed, although in instances where the ratio was >8, CFU counts indicated >2-log killing at or near the MIC.
- When the time-kill kinetics of ACX-362E were evaluated against *C. difficile* isolates MMX 5680 and BAA-1382, bactericidal activity was observed at the two later time points and at all three evaluated concentrations (MMX 5680) or the two highest doses (BAA-1382).
- Against *C. difficile* isolate BAA-1875, ACX-362E did not demonstrate the ≥3 $\log_{10}$ CFU/mL killing required for bactericidal activity, but bacterial levels were reduced by >2 $\log_{10}$ CFU at the 24- and 48-hr time points at 16X and 32X the MIC.
- Bactericidal activity for CDI is preferred so as to minimize the potential for relapse/recurrence in this disease.

REFERENCES

1) Evans, CT and Safdar, N. 2015. *Clin Infect Dis.* 60(52): 566 – 71.
2) Leffler, DA and Lamont, JT. 2015. *N Engl J Med.* 372: 1539 – 1548.
3) Jung KS,et al. 2010. *Gut Liver* 2010;4(3): 332-337.
4) Centers for Disease Control and Prevention. Antibiotic threats in the United States, 2013.
5) Torti, A, et al 2011. *Curr Enz Inhib.* 7:147 – 153.