PRTX-100 INHIBITS PLATELET PHAGOCYTOSIS *IN VITRO*

Chris Yatko, Christopher Herrem, Samia Siddiqui, Victor S. Sloan. (Intr. By David Kuter)

Protalex, Inc., New Hope, PA, USA

SUMMARY

In idiopathic thrombocytopenic purpura (ITP), autoantibodies bind to platelets which are then phagocytosed by monocytes/macrophages and removed by the reticuloendothelial system. PRTX-100 (Staphylococcal protein A) is being investigated for the treatment of ITP. We developed an *in vitro* flow cytometric assay to assess the effect of PRTX-100 on phagocytosis of opsonized platelets. Human monocytes were treated with PRTX-100 at concentrations of 250, 25, 2.5, 0.25 and 0.025 ng/ml and incubated with allogeneic human platelets that were opsonized with W6/32 IgG and labeled with a PerCP lipophilic dye. Afterwards, PE labeled anti-CD61 antibody was added to assess for surface bound platelets versus ingested platelets. Phagocytosis was determined by flow cytometric analysis. Percent phagocytosis was calculated as the fraction of ingested platelets (PerCP+/CD61-) from the total PerCP population (PerCP+/CD61-) + (PerCP+/CD61+) within the gated monocyte population. PRTX-100 was found to inhibit the phagocytosis of opsonized platelets in a dose-dependent fashion. Phagocytosis was dependent on opsonization with W6/32, indicating a role of Fc receptors on the effector monocytes. Since prevention of platelet phagocytosis is an important treatment goal in ITP, PRTX-100 is currently under investigation as a possible therapy for patients with ITP.

INTRODUCTION

PRTX-100

- Purified native Staphylococcal protein A (SpA), a 47 kDa protein.
- Five homologous 58-61 amino acid domains.
- Contains two distinct types of Ig binding activities, an Fc_{γ} binding and a V_H3 restricted Fab binding

ITP

- An autoimmune disorder characterized by low platelet counts
- IgG auto-antibodies are directed primarily towards the platelet surface marker glycoprotein IIb/IIIa complex
- Auto-antibody opsonized platelets are destroyed via Fc_{γ} receptor mediated phagocytosis by macrophages.

Given that PRTX-100 binds to 100% of human monocytes, we hypothesize that treatment of human monocytes/macrophages with PRTX-100 would result in a reduction in phagocytosis of opsonized platelets.

PURPOSE

To assess the effect of PRTX-100 on phagocytosis of platelets in an *in vitro* flow cytometric assay.

METHODS

Platelet Phagocytosis assay

Culture monocytes for 6 days
↓
48 hour PRTX-100 treatment
↓
1 hour coculture with W6/32 opsonized PerCP labeled platelets
↓
Measure phagocytosis by flow cytometry

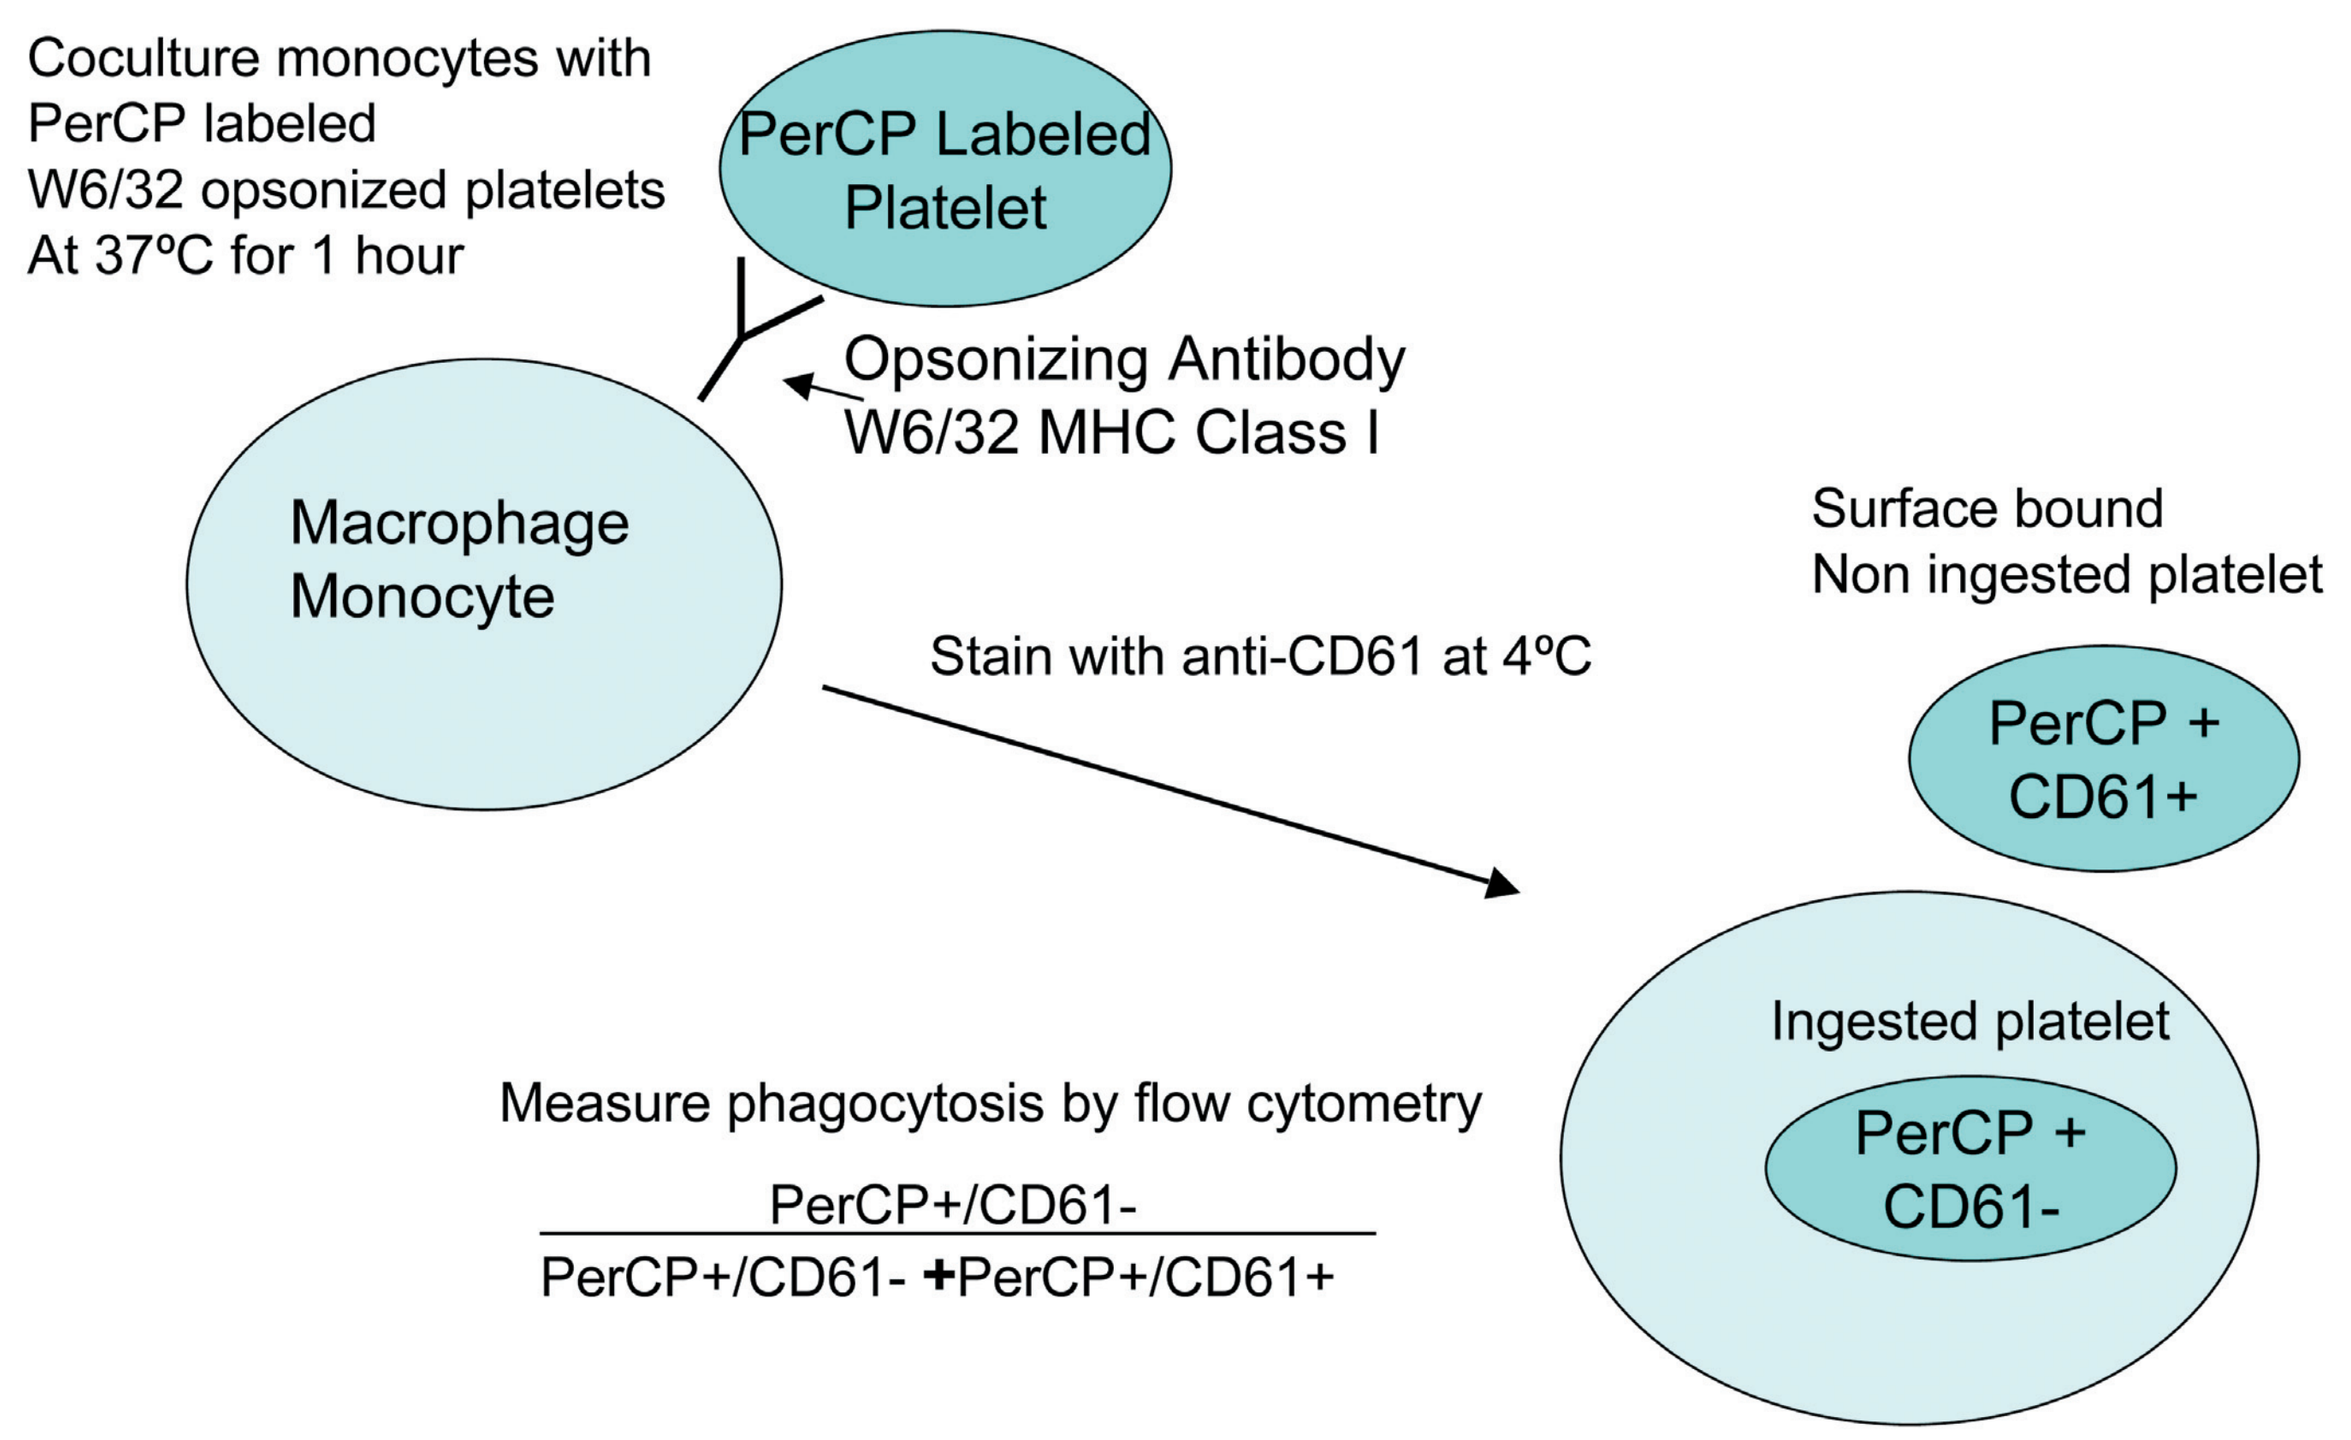
Coculture monocytes with PerCP labeled W6/32 opsonized platelets At 37°C for 1 hour
PerCP Labeled Platelet
Opsonizing Antibody W6/32 MHC Class I
Macrophage Monocyte
Stain with anti-CD61 at 4°C
Surface bound Non ingested platelet
PerCP + CD61+
Ingested platelet
PerCP + CD61-
Measure phagocytosis by flow cytometry
PerCP+/CD61-
PerCP+/CD61- +PerCP+/CD61+

RESULTS

Human monocytes gated for analysis verified by CD14 expression

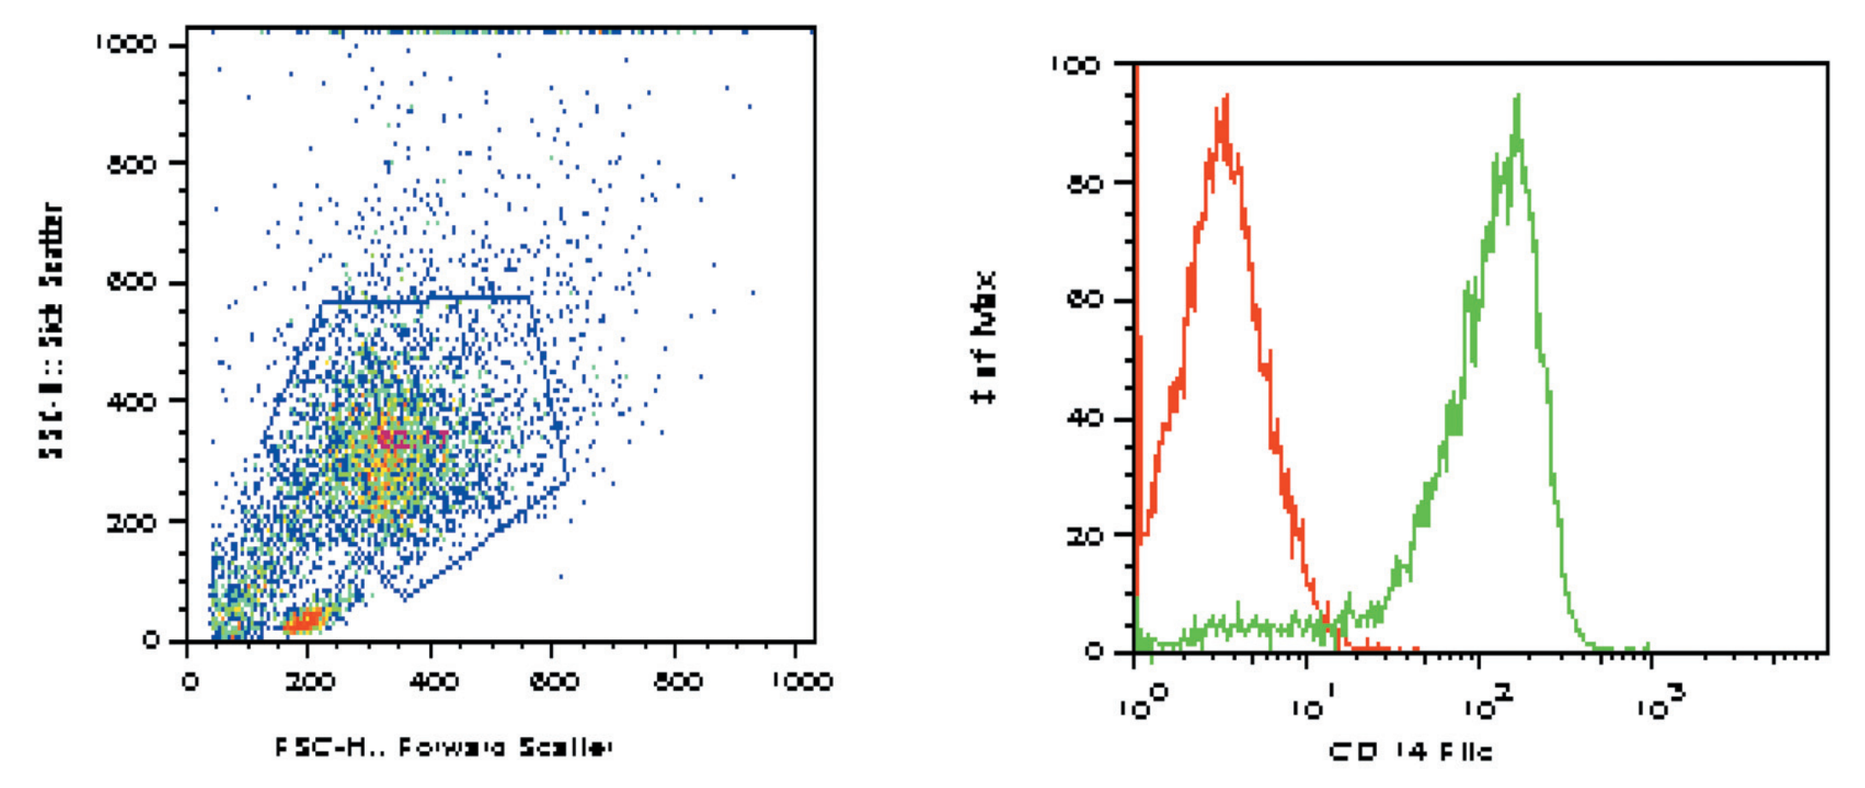
FSC-H:: Forward Scatter
CD 14 FITC

Phagocytosis of platelets is dependent on W6/32 opsonization

Opsonized platelets 37°C	Non-opsonized platelets 37°C	Opsonized platelets 4°C

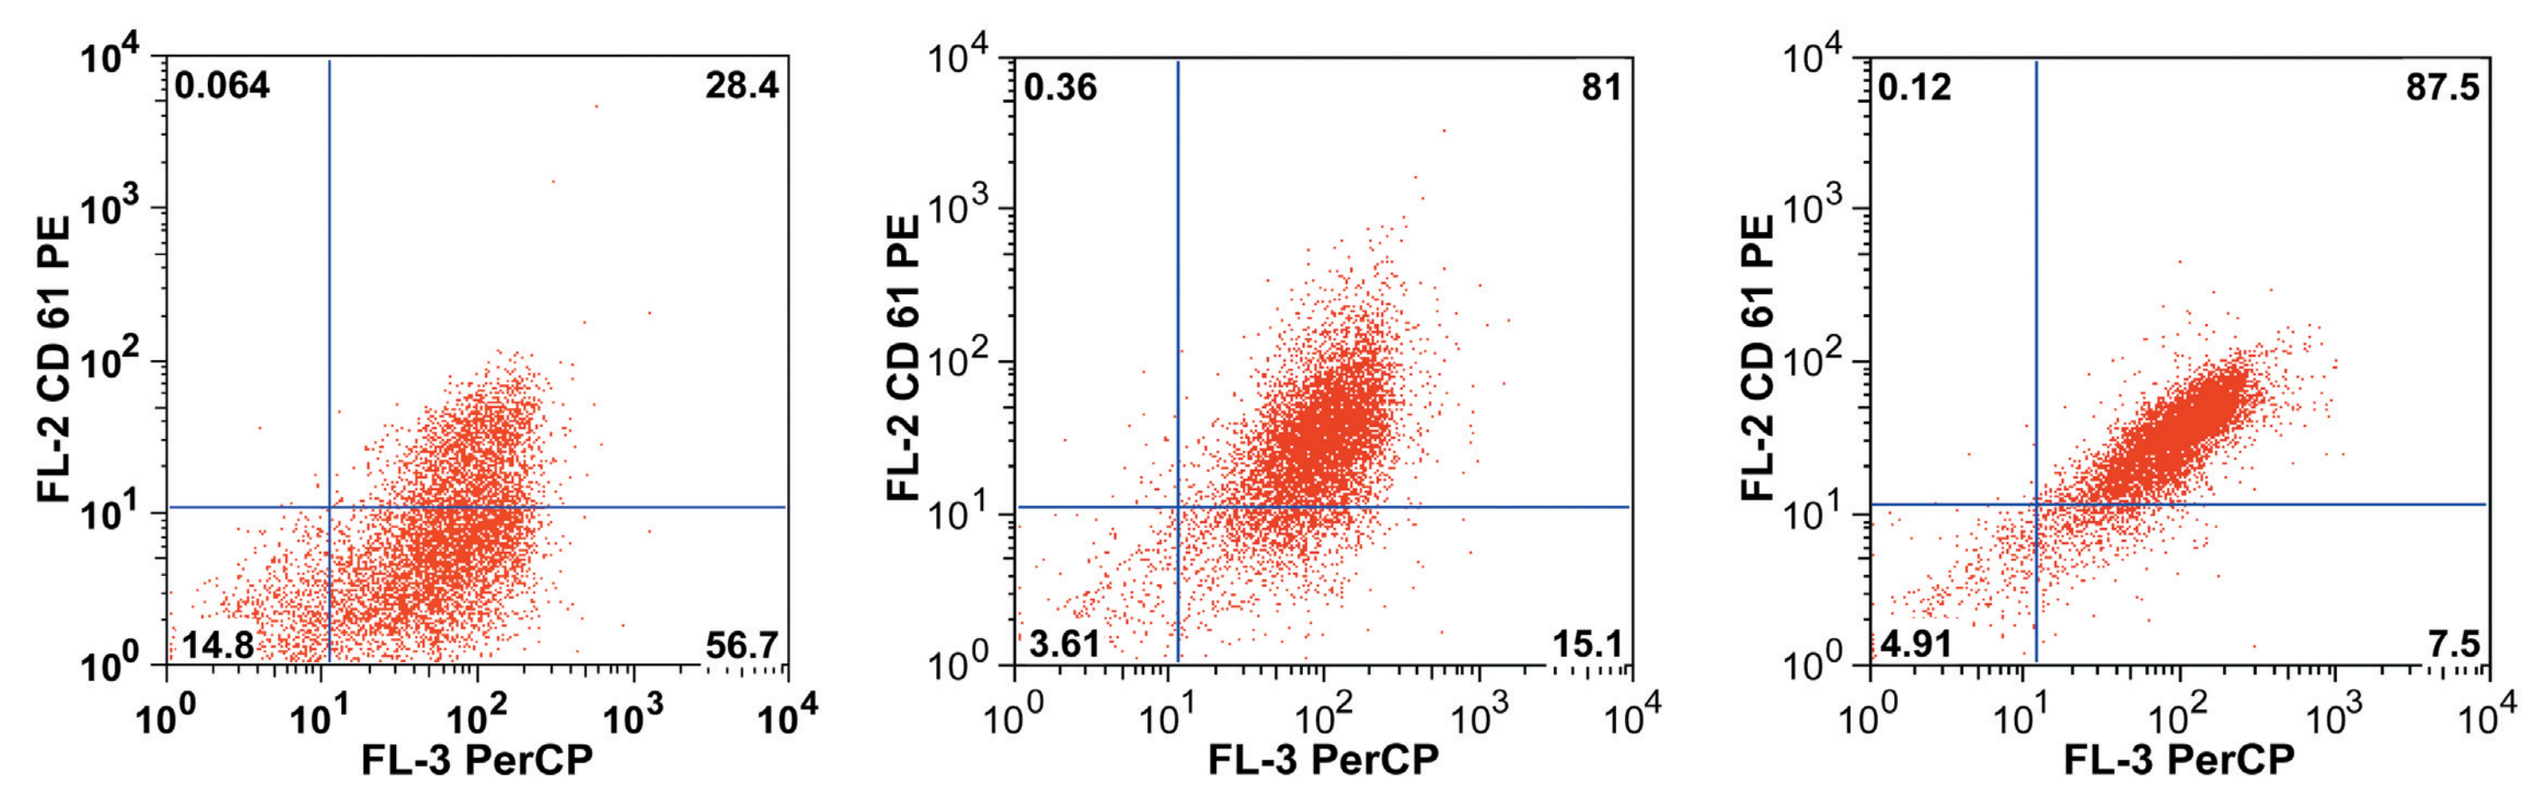
FL-2 CD 61 PE
FL-3 PerCP
0.064
28.4
14.8
56.7
0.36
81
3.61
15.1
0.12
87.5
4.91
7.5

Flow cytometric analysis of the effect of PRTX-100 on phagocytosis of opsonized platelets

PRTX-100 250 ng/ml

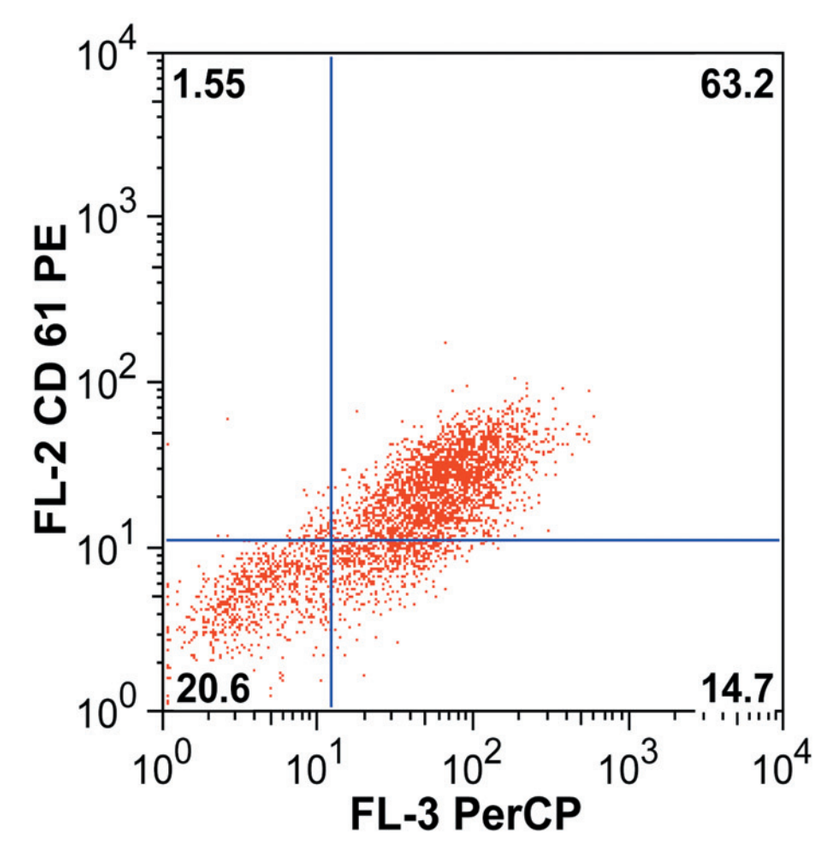
FL-2 CD 61 PE
FL-3 PerCP
1.55
63.2
20.6
14.7

PRTX-100 25 ng/ml

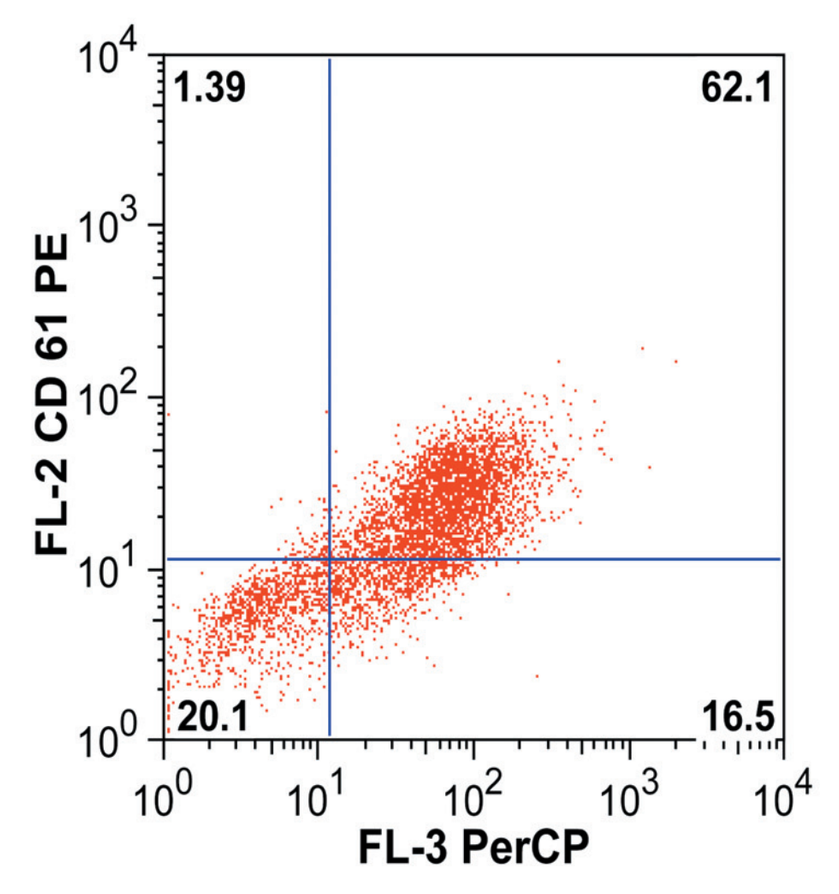
FL-2 CD 61 PE
FL-3 PerCP
1.39
62.1
20.1
16.5

PRTX-100 2.5 ng/ml

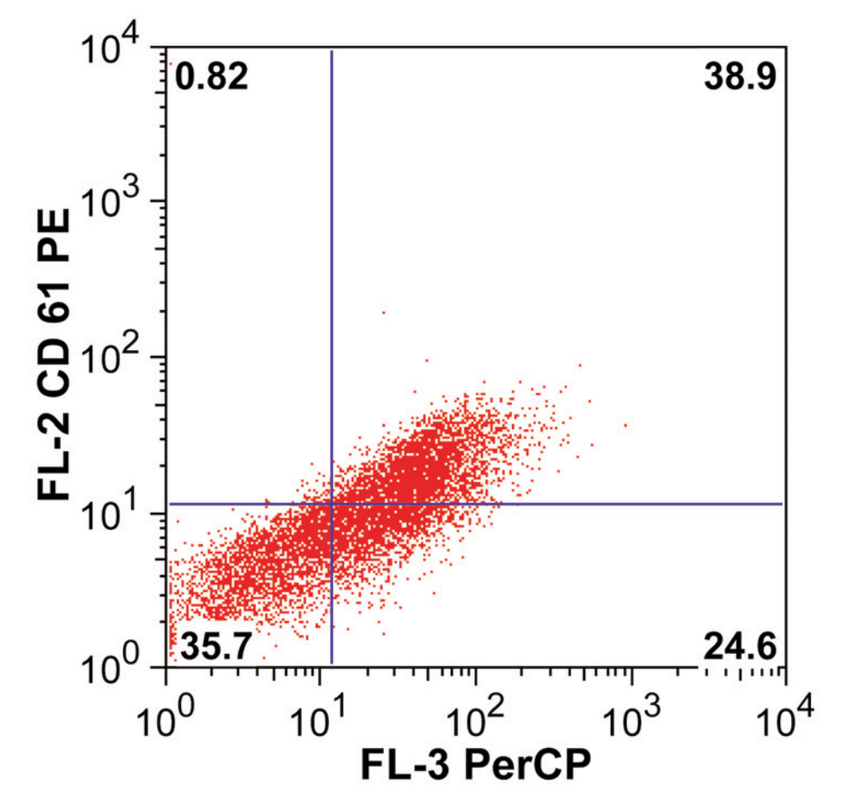
FL-2 CD 61 PE
FL-3 PerCP
0.82
38.9
35.7
24.6

PRTX-100 0.25 ng/ml

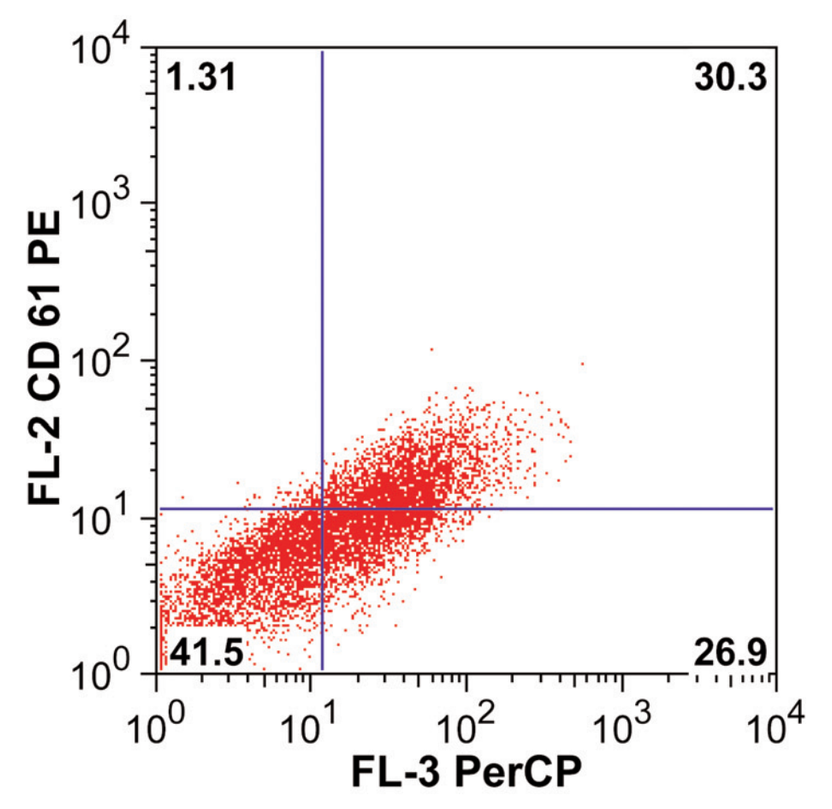
FL-2 CD 61 PE
FL-3 PerCP
1.31
30.3
41.5
26.9

PRTX-100 0.025 ng/ml

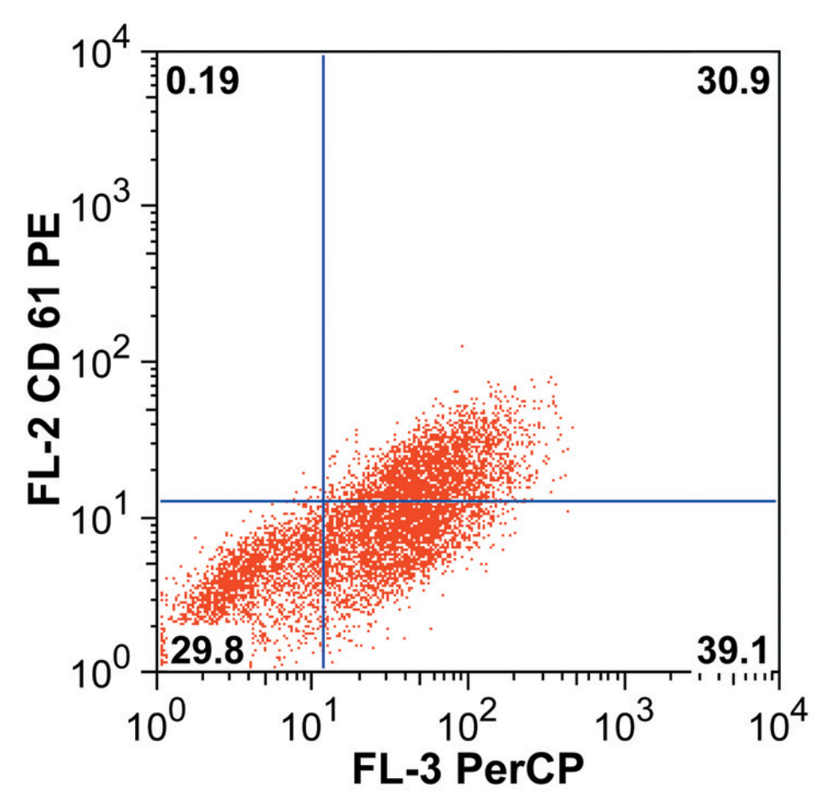
FL-2 CD 61 PE
FL-3 PerCP
0.19
30.9
29.8
39.1

Effect of PRTX-100 on *in vitro* platelet phagocytosis

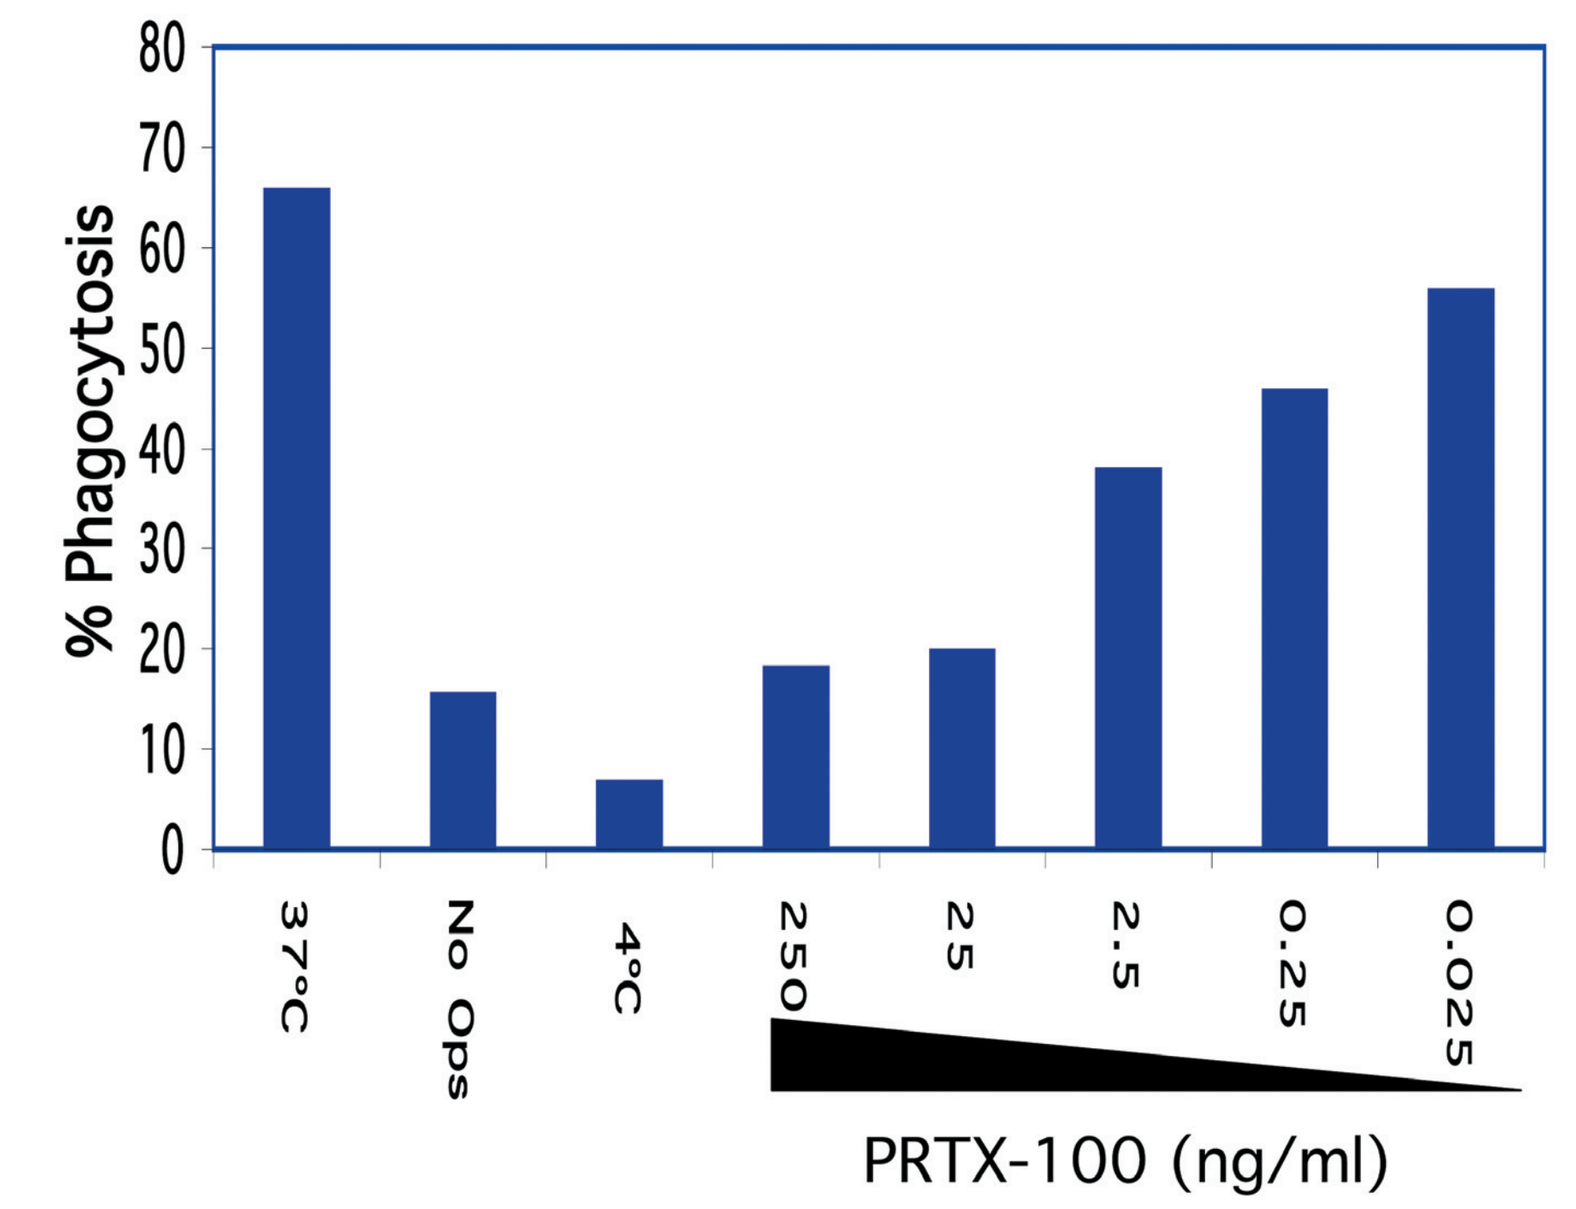
% Phagocytosis
37°C
No Ops
4°C
250
25
2.5
0.25
0.025
PRTX-100 (ng/ml)

CONCLUSIONS

PRTX-100 inhibits the phagocytosis of W6/32 opsonized platelets by human monocytes. Phagocytosis of W6/32 opsonized platelets in the absence of PRTX-100 was 66% while that of non-opsonized platelets was 15%. These data demonstrate that the observed phagocytosis was antibody dependent and presumably involves Fc receptors on the monocytes.

Phagocytosis in the presence of PRTX1-100 at concentrations of 250, 25, 2.5, 0.25 and 0.025 was 18%, 20%, 38%, 46%, and 56%, respectively. These date demonstrate a dose dependent effect of the ability of PRTX-100 to inhibit platelet phagocytosis by human monocytes.

ACKNOWLEDGMENTS

We would like to thank Dr. John Semple of the University of Toronto for his helpful discussions on the development of this work.

DISCLOSURE

The authors are full-time employees of Protalex, Inc.; the authors hold stock and/or options at Protalex, Inc.; the research presented was funded entirely by Protalex, Inc.