Medical Affairs:
Deb Mathews
dmathews@syntheticbiologics.com

Synthetic BIOLOGICS

Presented by:
John F. Kokai-Kun
jkokai-kun@syntheticbiologics.com

Investor Relations:
Vincent Perrone
vperrone@syntheticbiologics.com

The Human Microbiome

The Human Microbiome

A collection of microorganisms living symbiotically in and around the human body

Human body has 10 times as many microbial cells as human cells

Human Microbiome > 1,000,000 Genes

Human Genome 23,000 Genes

99% of genes in the body are Microbial, NOT Human

Diseases Directly Influenced by the Gut Microbiome

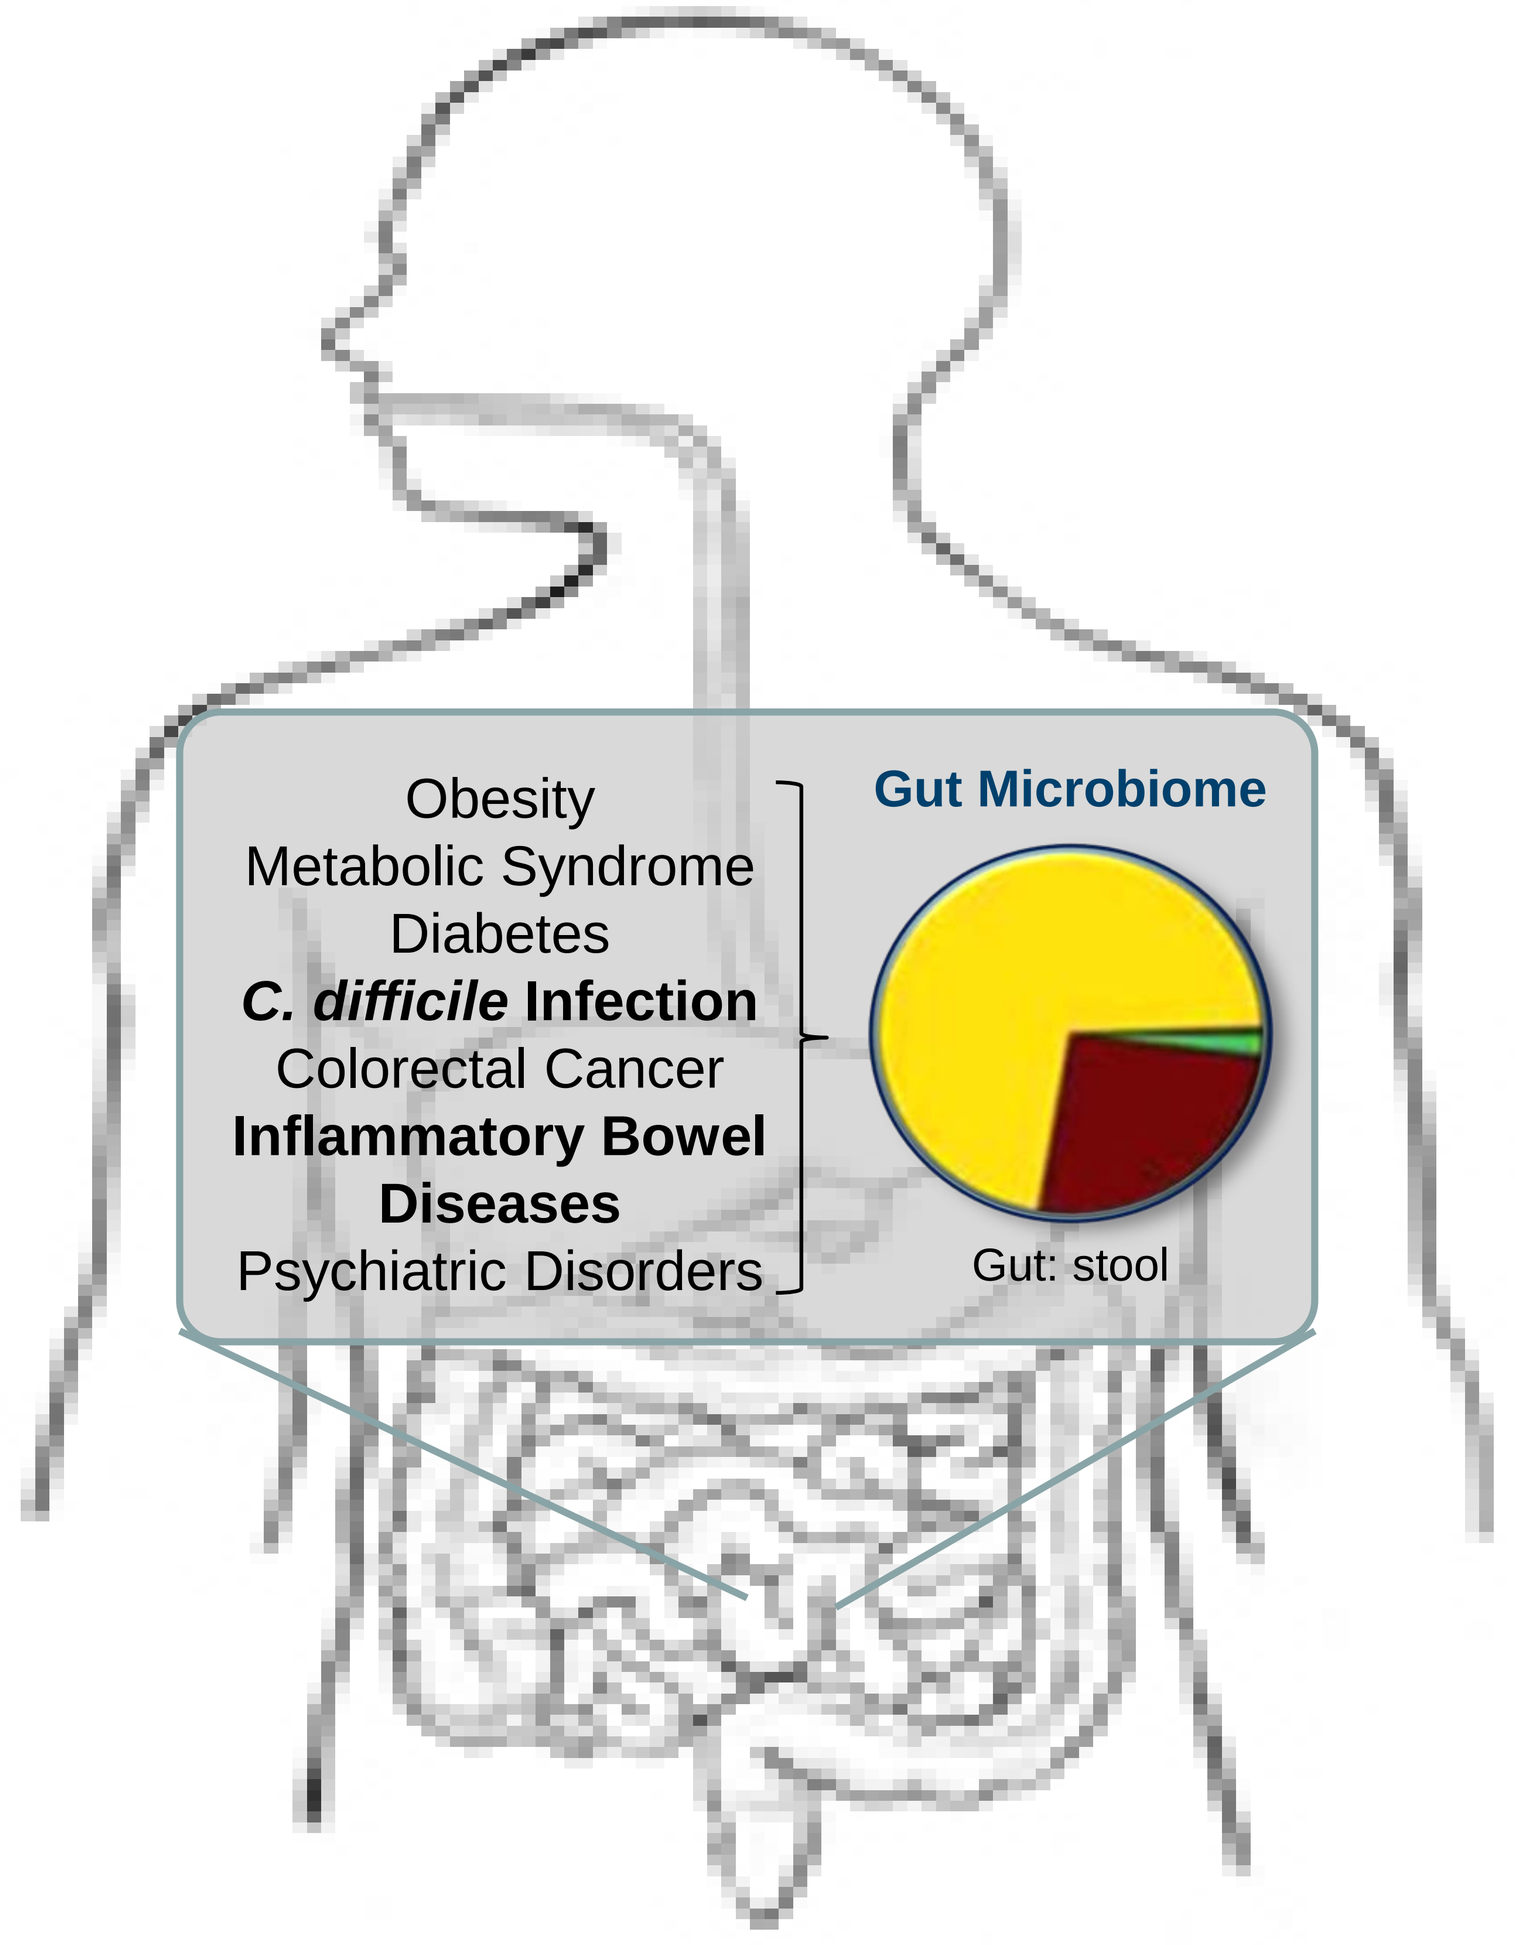
Obesity
Metabolic Syndrome
Diabetes
C. difficile Infection
Colorectal Cancer
Inflammatory Bowel Diseases
Psychiatric Disorders
Gut Microbiome
Gut: stool

Synthetic Biologics, Inc.

Advancing Gut Microbiome Therapeutics

Synthetic Biologics is developing cutting-edge therapeutics to treat pathogen-specific diseases

- A late-stage clinical stage company developing therapeutics to protect the gut microbiome and restore the health of patients while targeting pathogen-specific diseases. Founded in 2001, Synthetic Biologics, Inc. trades on the NYSE MKT, LLC under the symbol "SYN". Located in Rockville, MD USA
- The Company's lead candidates poised for Phase 3 development are: (1) SYN-010 which is intended to reduce the impact of methane producing organisms in the gut microbiome to treat an underlying cause of irritable bowel syndrome with constipation (IBS-C), and (2) SYN-004 (ribaxamase) which is designed to protect the gut microbiome from the effects of certain commonly used intravenous (IV) beta-lactam antibiotics for the prevention of *C. difficile* infection (CDI), antibiotic-associated diarrhea (AAD) and the emergence of antimicrobial-resistance (AMR).

Product Pipeline

Therapeutic Area / Product Candidate	Discovery	Preclinical	Phase 1	Phase 2	Phase 3
Irritable Bowel Syndrome-Constipation (IBS-C) **SYN-010 [C]** ***Oral modified-release lovastatin lactone***					
Prevention of *C. difficile* infection (CDI) **SYN-004 (ribaxamase)** ***Oral enzyme to degrade IV β-lactam Antibiotics***					
Prevention of CDI SYN-007 *Oral enzyme to degrade orally administered Beta-lactam Antibiotics*					
Prevention of CDI SYN-006 *Oral enzyme to degrade IV Carbapenem Antibiotics*					
Prevention/Treatment of Pertussis (Whooping Cough) SYN-005 [I,T] *Monoclonal antibodies*					
Phenylketonuria (PKU) SYN-200 [I] *Biotherapeutics*					
Preserve gut microbiome and barrier/Treat Inflammation SYN-020 *Oral intestinal alkaline phosphatase*					

Completed
Planned

C - Cedars-Sinai Medical Center collaboration, I - Intrexon Corporation collaboration, T - The University of Texas at Austin collaboration

SYN-010

A proprietary modified-release formulation of lovastatin lactone

The Role of Methane in IBS-C

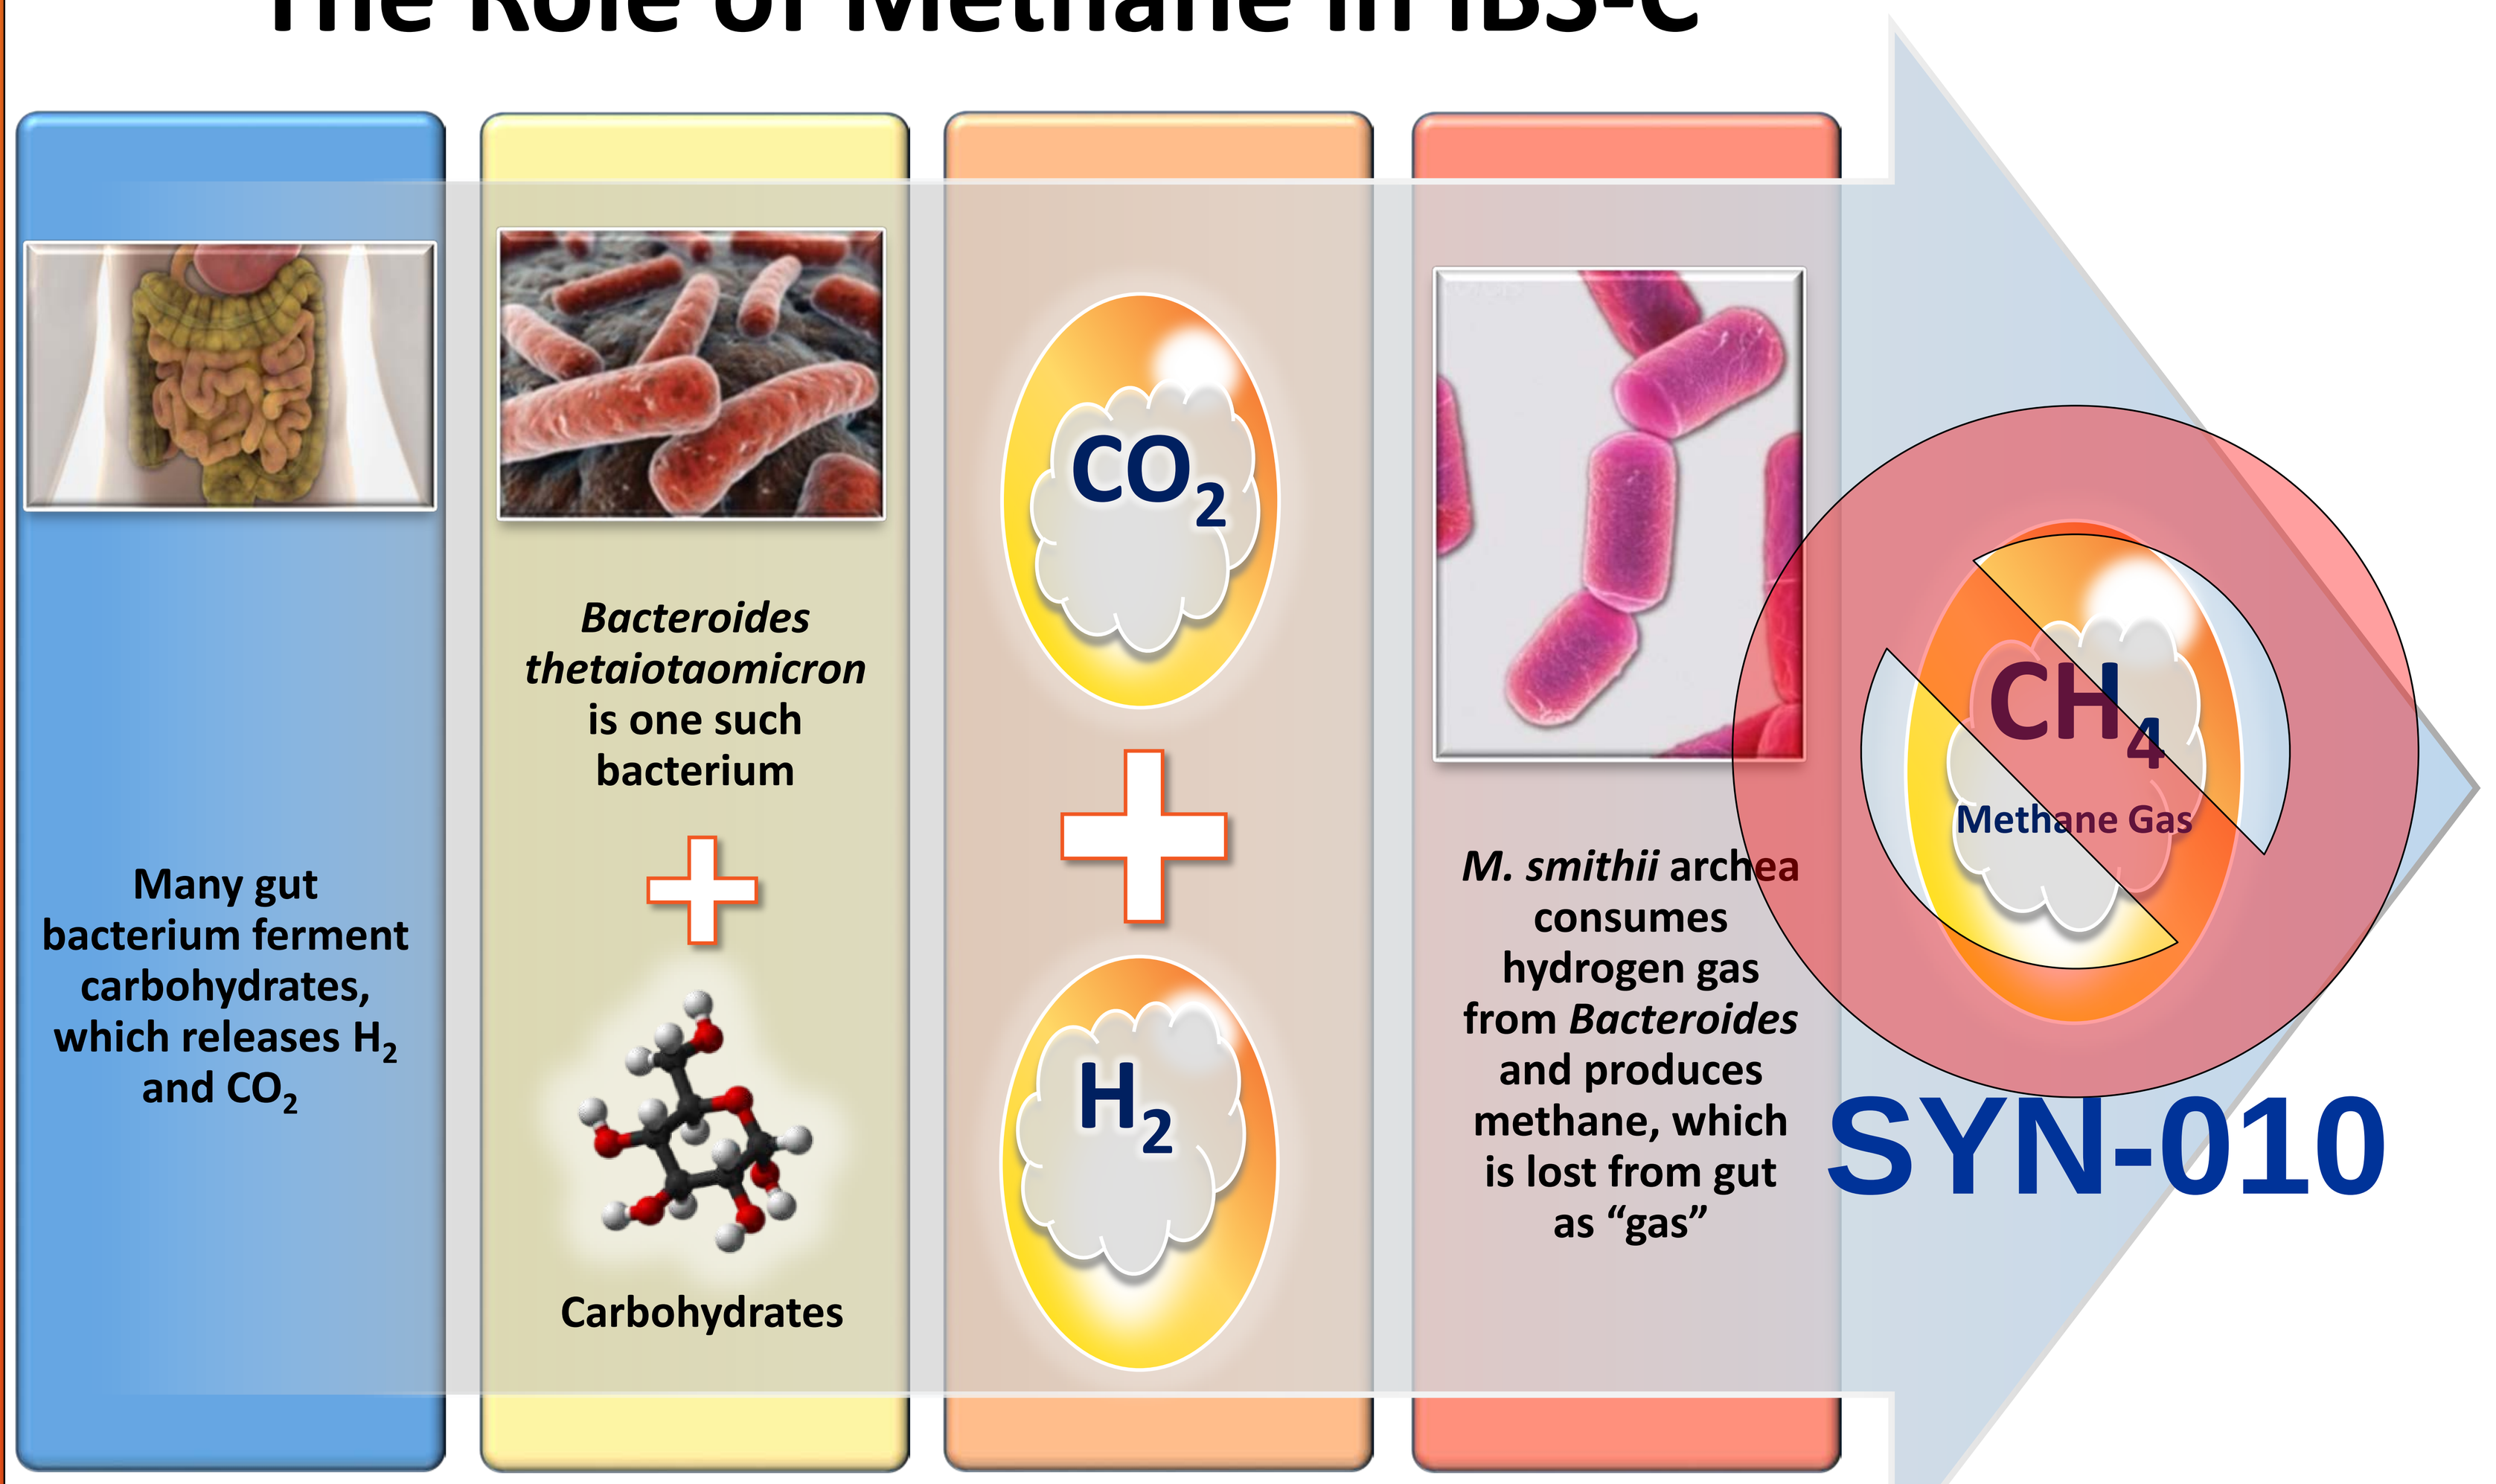
Many gut bacterium ferment carbohydrates, which releases H_2 and CO_2
Bacteroides thetaiotaomicron is one such bacterium
Carbohydrates
CO_2
H_2
M. smithii archea consumes hydrogen gas from Bacteroides and produces methane, which is lost from gut as "gas"
CH_4
Methane Gas
SYN-010

Modulating *M. smithii* archaea to reduce methane and alleviate the symptoms of irritable bowel syndrome with constipation

SYN-004 (ribaxamase)

Degrading Residual IV β-lactam Antibiotics May Prevent *C. difficile* infection, antibiotic-associated diarrhea and antimicrobial resistance

SYN-004 to be orally co-administered with intravenous β-lactam antibiotics

- Ribaxamase is designed to neutralize active β-lactam antibiotics in the GI tract and thus eliminate selective pressure

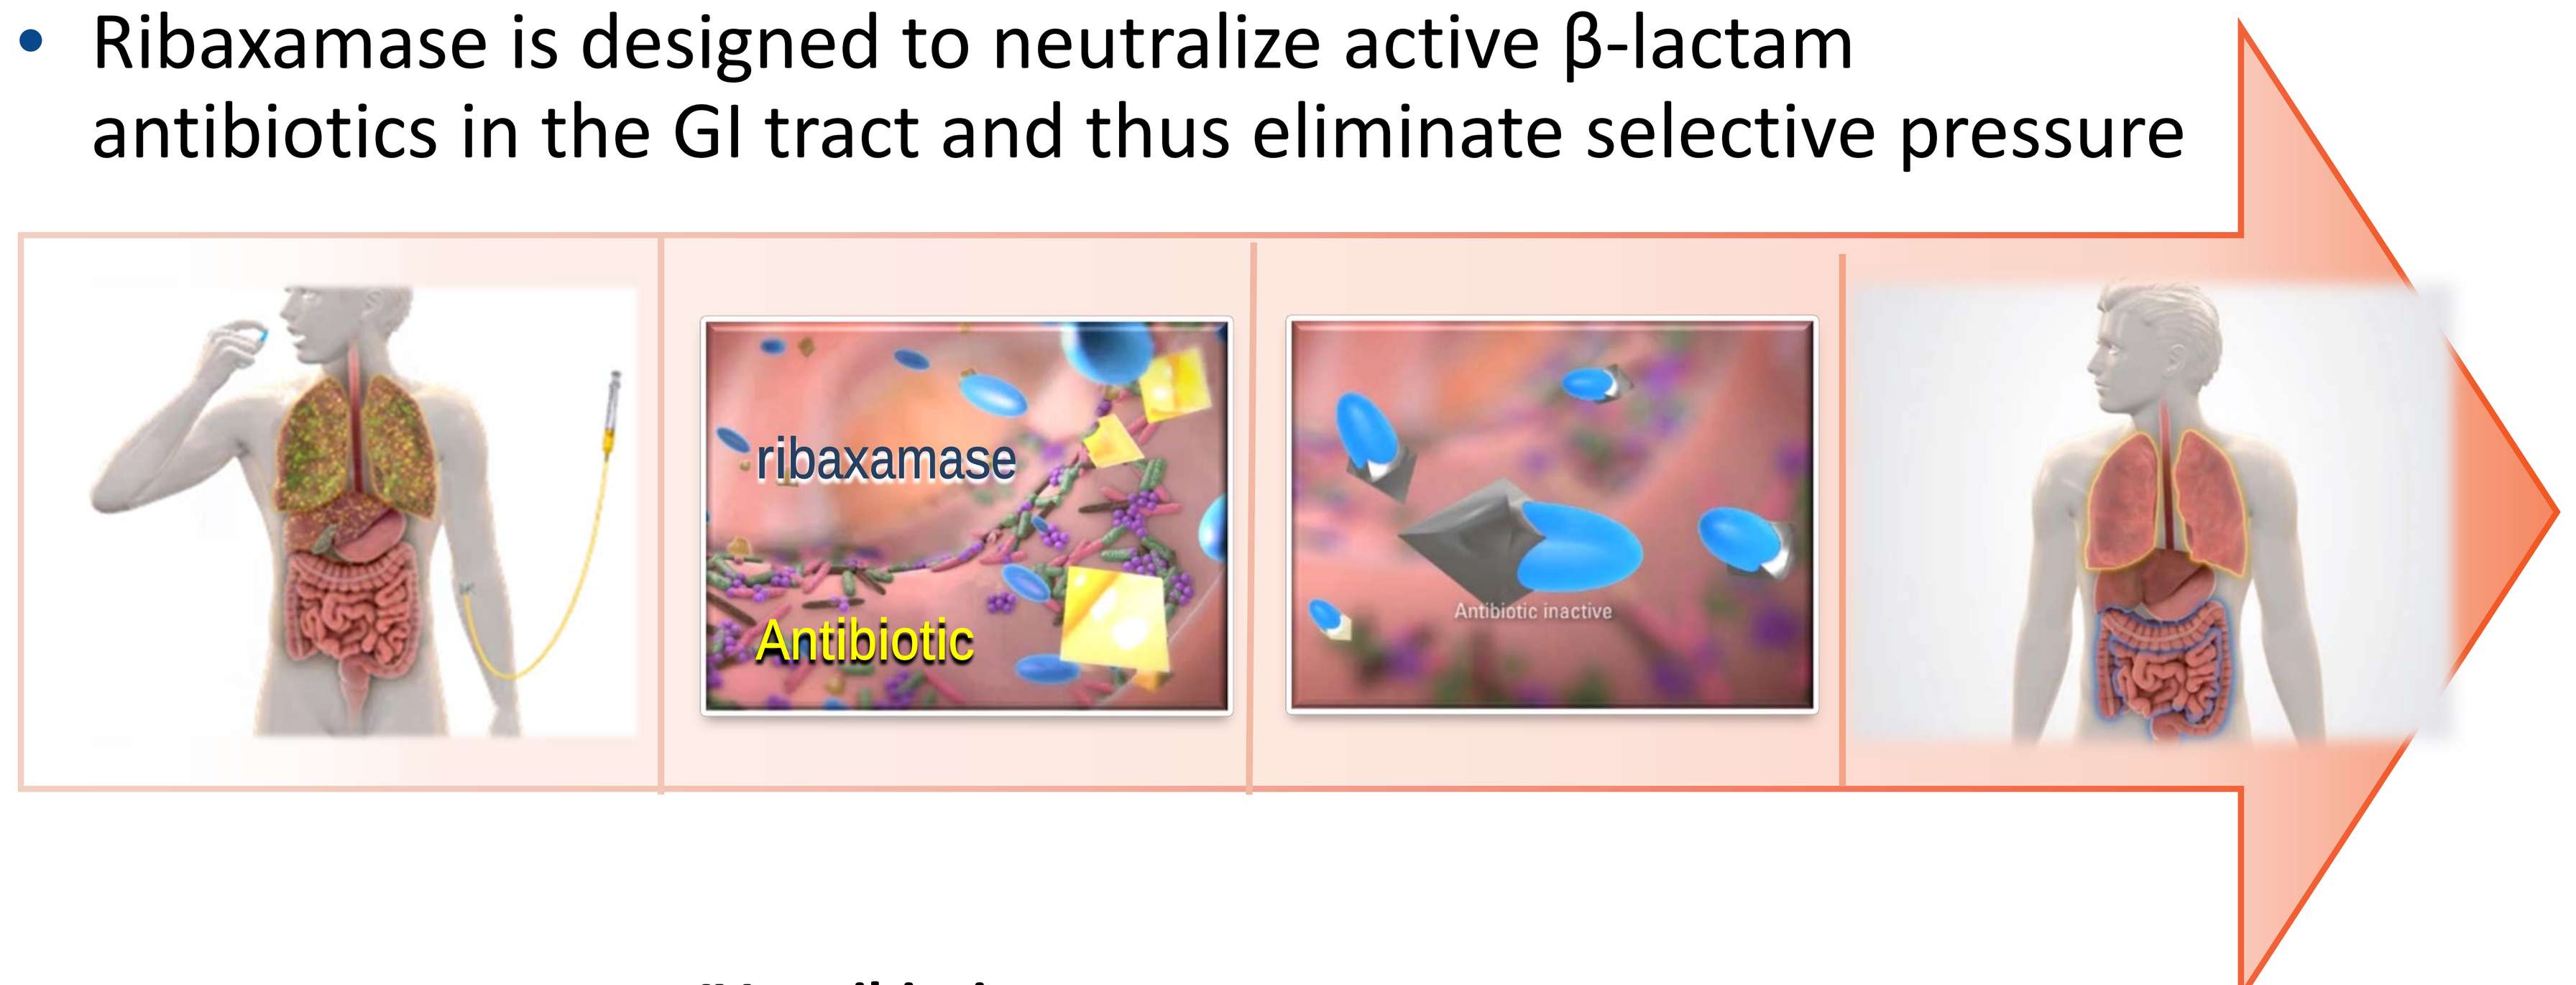
ribaxamase
Antibiotic
Antibiotic inactive

Ribaxamase is an oral enzyme tablet to be co-administered with IV β-lactam antibiotics.

IV antibiotics can upset the natural balance of the gut microbiome, killing "good" bacteria, allowing for the overgrowth of *C. difficile.*

Ribaxamase is intended to remain in the GI tract and neutralize IV antibiotics (black), and to protect the natural balance of the gut microbiome.

Co-administration of ribaxamase is intended to allow IV antibiotic to treat the primary infection while protecting the gut microbiome

Development Milestones

Therapeutic Area / Product Candidate	Timeline
IBS-C – SYN-010:	
Phase 2 (1st study; acute, placebo-controlled)	√ **2Q 2015 – Initiated Phase 2** √ **4Q 2015 – Reported Phase 2 topline data**
Phase 2 (2nd study; extension, SYN-010 42mg)	√ **4Q 2015 – Initiated Phase 2** √ **1Q 2016 – Reported Phase 2 topline data**
Held End of Phase 2 Meeting with FDA	√ **Summer 2016**
Pivotal Phase 2b/3 trial	**2017 – Initiate Phase 2b/3 trial**
CDI/AAD Prevention – SYN-004 (ribaxamase):	
Phase 1a/1b	√ **1Q 2015 – Supportive topline Phase 1b results** √ **1Q 2015 – Supportive Phase 1a/1b PK data**
Phase 2a open-label (1st ileostomy study; ceftriaxone)	√ **1Q 2015 – Initiated Phase 2a** √ **4Q 2015 – Reported supportive Phase 2a topline data**
Phase 2a open-label (2nd ileostomy study; ceftriaxone + PPI)	√ **2Q 2015 – Initiated Phase 2a** √ **2Q 2016 – Report Phase 2a topline data**
Phase 2b proof-of-concept (double-blind, placebo-controlled)	√ **3Q 2015 – Initiated Phase 2b trial** √ **1Q 2017 – Announce topline data from Phase 2b**
Hold End of Phase 2 Meeting with FDA	**2H 2017**
Pivotal Phase 3 trial(s)	**1H 2018 – Initiate Phase 3 trial(s)**

Synthetic Biologics, Inc.