Clinical Development for Monoclonal Antibody Therapy to Treat and Prevent Neonatal Pertussis

Ekaterina Vert-Wong*, Joseph Sliman, J. Andrew Bristol, Michael Kaleko, Olivia Coughlin, Jennifer Maynard, Roman Wolf

Synthetic Biologics, Inc., Rockville, MD, USA

kvert-wong@syntheticbiologics.com
240-676-2665

Abstract

OBJECTIVE: Pertussis is a significant public health problem despite near-universal vaccination and continues to cause up to 300,000 deaths worldwide each year, primarily among unvaccinated infants. Antibiotic therapy is ineffective, presumably due to lack of clearance of pertussis toxin (PTx), a major virulence factor of *B. pertussis* bacterium.

DESIGN AND METHODS: SYN-005 was engineered to treat and prevent pertussis in newborns and infants to decrease morbidity and mortality, reduce pediatric intensive care unit (PICU) admissions and shorten hospitalization. SYN-005 is a cocktail of two uniquely potent and synergistic humanized mAbs, designed to neutralize pertussis toxin and manufactured by recombinant CHO cell lines. SYN-005 was evaluated in mouse and baboon pertussis models. Clinical development of SYN-005 is planned as a single dose, IM injection to be administered at birth to newborns in geographic areas where pertussis is endemic, once the nonclinical safety and pharmacology profile is established.

RESULTS AND CONCLUSION: In the animal studies, compared to the untreated controls, treatment with SYN-005 rapidly blunted the rise in white blood cell (WBC) count, accelerated bacterial clearance from the nasopharynx, and in baboons, shortened the course of coughing. No signs of toxicity were noted. The data from nonclinical studies and the approach for clinical development will be presented. Synthetic Biologics intends to pursue clinical development of SYN-005 once safety is established. A dose-ranging study in the newborn target population is proposed that will measure serum levels of SYN-005 over time using the WHO standard as a reference. Efficacy and safety assessments will be performed with the goal of determining a trend towards protection.

Background

SYN-005 is a combination of two uniquely potent and synergistic antibodies designed to bind and neutralize pertussis toxin. 11E6 binds to the toxin's S2 and S3 subunits and blocks toxin binding to its target cells. 1B7 binds to both the S1 and S4 subunits, prevents their dissociation within the cell, and blocks the toxic ADP-ribosylase activity of the S1 subunit. The antibodies may be used individually or in combination (referred to here as SYN-005) to treat infants with active pertussis infection. Furthermore, given that the vast majority of the world's children live in circumstances where advanced pediatric critical care services are unavailable, the antibodies may be used to prevent infection in vulnerable newborns.

Preclinical Data

Prophylactic Treatment with Humanized Abs Protects Mice Against Pertussis

Mice were treated with the 1B7 and 11E6 antibodies (20 ug total dose) via IP injection 2 hrs prior to infection with 5x10⁶ CFU *B. pertussis* D420 bacteria. Leukocytes, body weight, and bacterial colonization of the lungs were evaluated 10 days later.

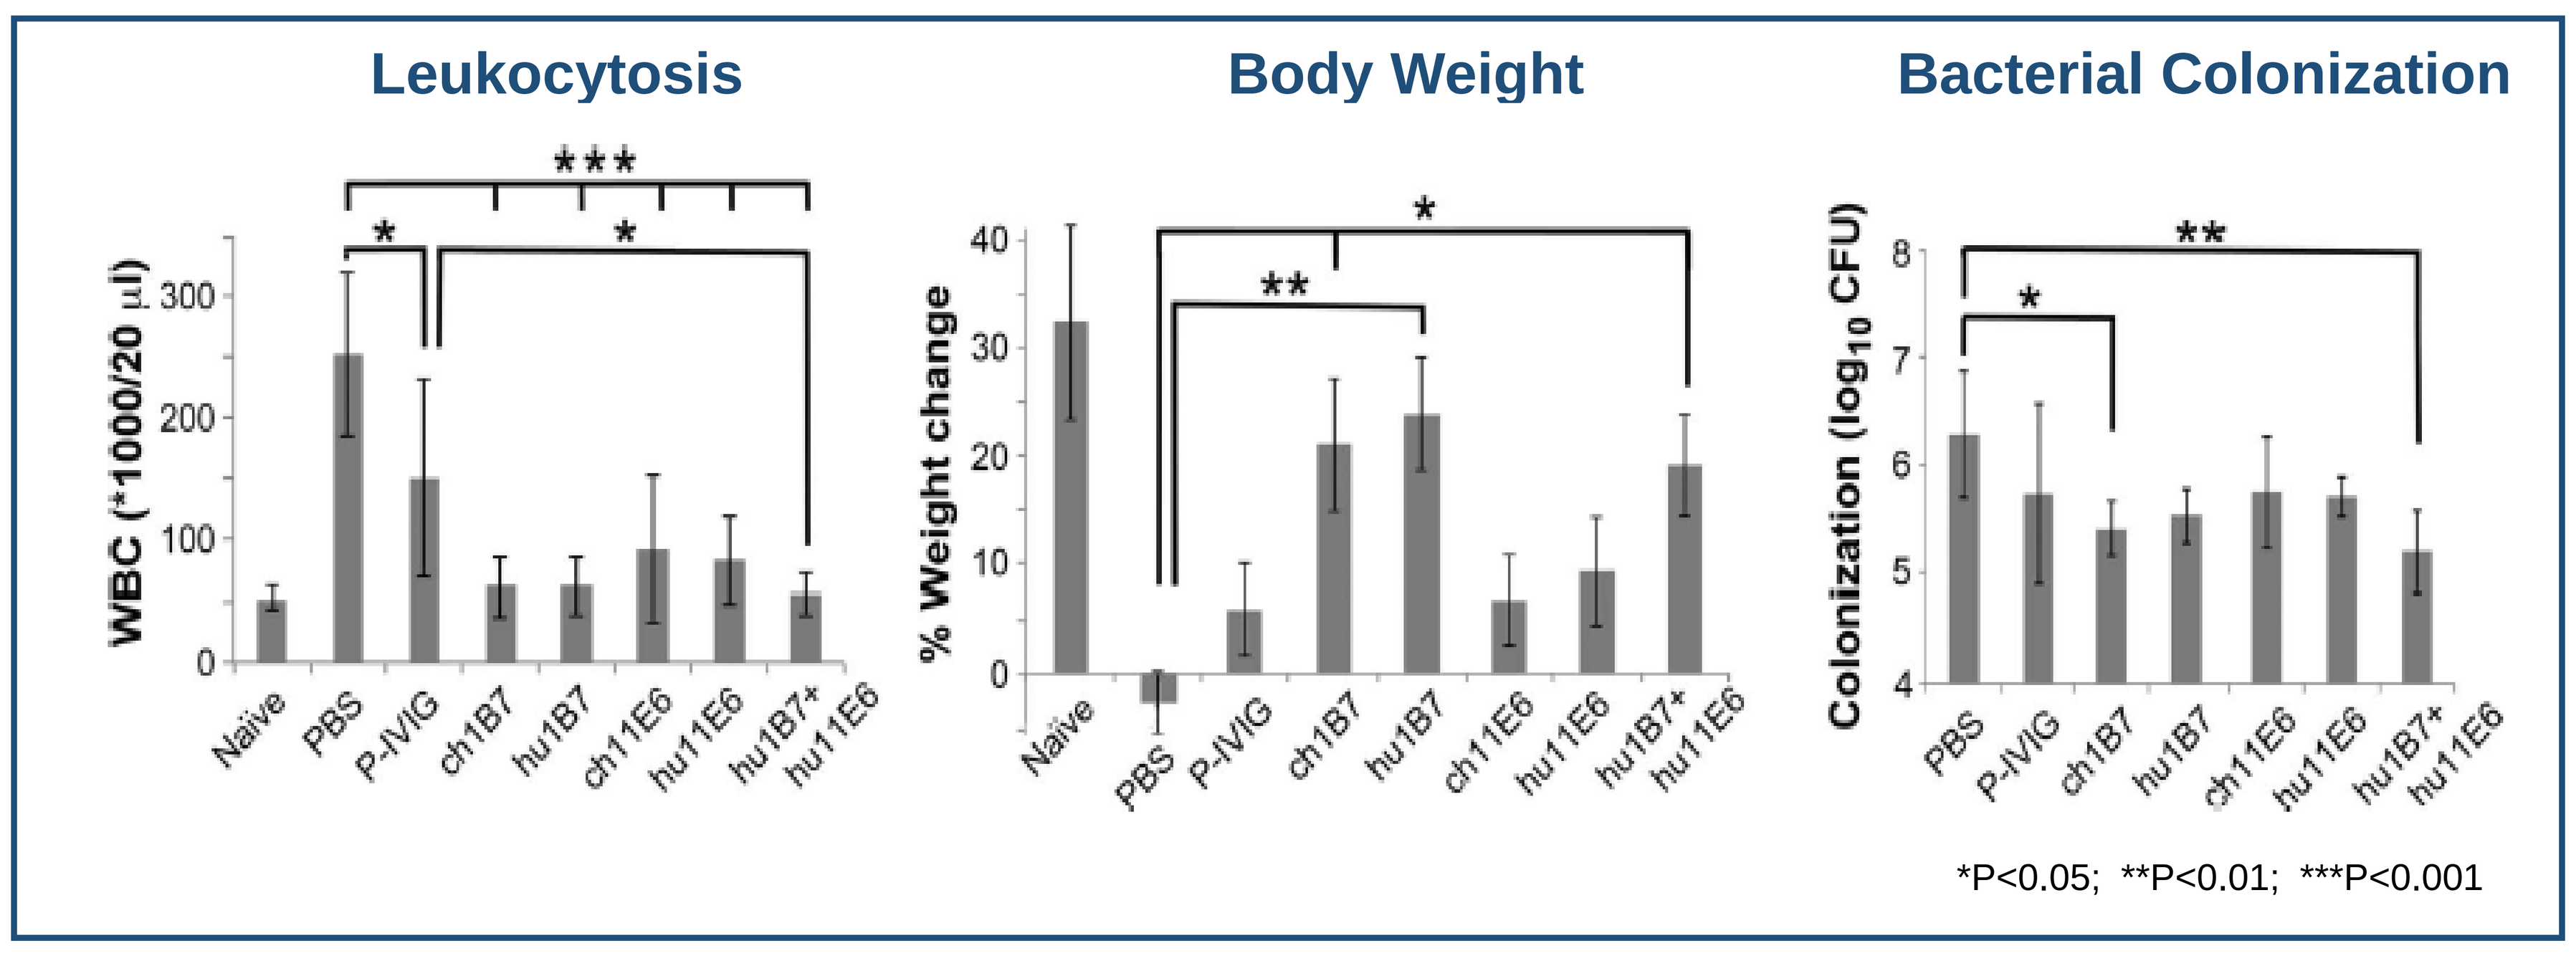
Leukocytosis
Body Weight
Bacterial Colonization
WBC (*1000/20 µl)
% Weight change
Colonization (log10 CFU)
Naïve
PBS
P-IVIG
ch1B7
hu1B7
ch11E6
hu11E6
hu1B7+ hu11E6
*P<0.05; **P<0.01; ***P<0.001

The individual antibodies and the binary combination mitigated disease with respect to white blood cell (WBC) count and *B. pertussis* lung colonization. Antibody treatment enabled the mice to gain weight. The humanized antibodies were as efficacious as previous chimeric versions and more effective than P-IVIG, a hyperimmune globulin previously used in human clinical trials.

Therapeutic Efficacy of Humanized Abs in Pertussis-Infected Baboons

Weanling baboons (n=8) were infected with 10^9-10^{10} CFU of *B. pertussis* D420 bacteria on Day 0 and four were treated on Day 3 with both hu1B7 and hu11E6 (20 mg/kg of each IV).

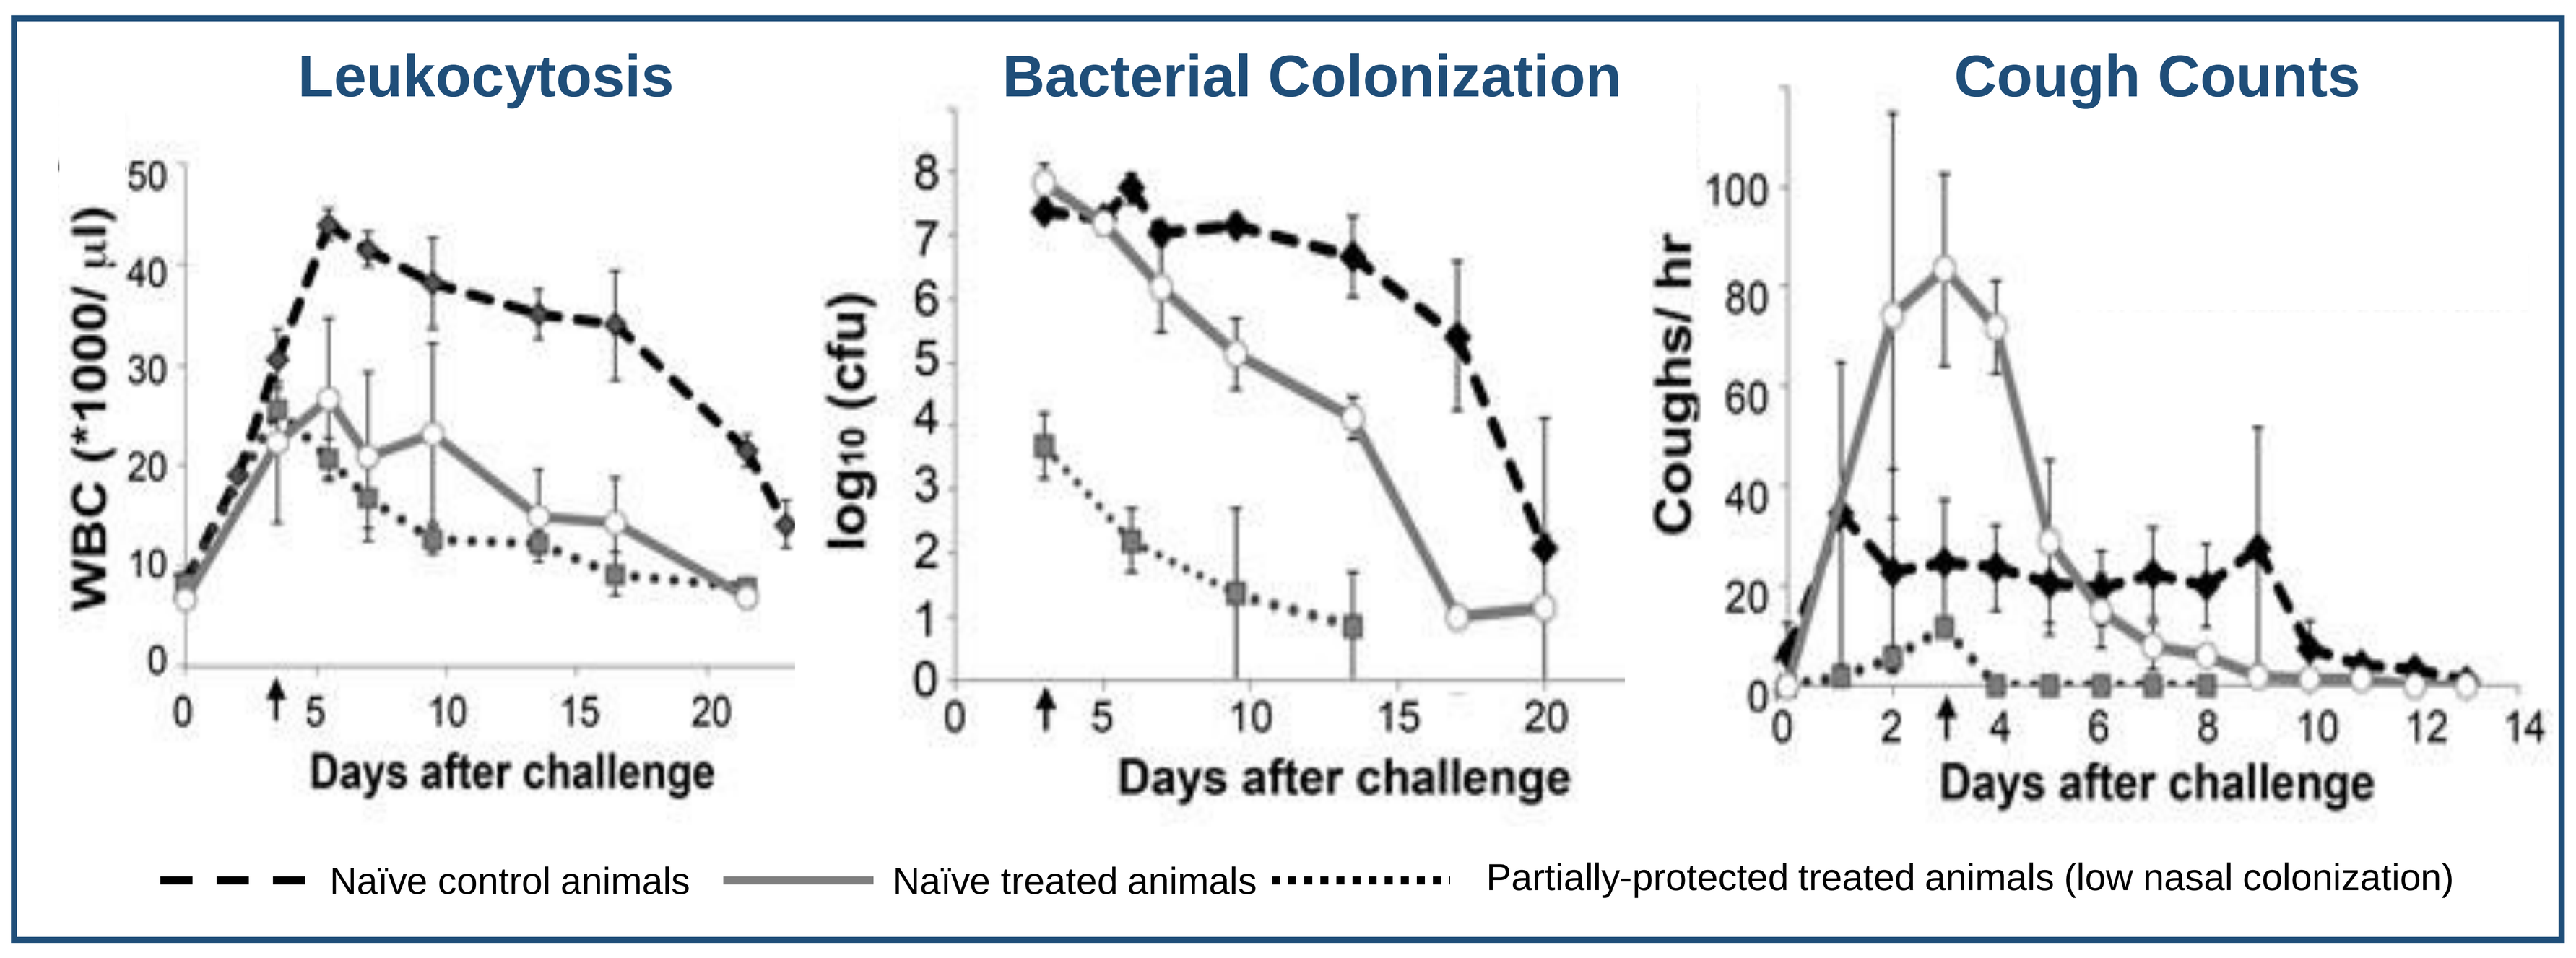
Leukocytosis
Bacterial Colonization
Cough Counts
WBC (*1000/ µl)
log10 (cfu)
Coughs/ hr
Days after challenge
Naïve control animals
Naïve treated animals
Partially-protected treated animals (low nasal colonization)

SYN-005 treatment reversed leukocytosis, accelerated bacterial clearance, and alleviated coughing. These data indicate that SYN-005 neutralizes pertussis toxin *in vivo* which, in a patient, may lead to immune recovery to fight the infection and may prevent the potentially fatal complication of pulmonary hypertension.

Data published in Nguyen et al., Kaleko, and Maynard (2015). "A cocktail of humanized anti-pertussis toxin antibodies limits disease in murine and baboon models of whooping cough". Science Translational Medicine, Dec 2;7(316):316ra195. doi: 10.1126/scitranslmed.aad0966.

Antibody 1B7 Undergoing Testing in Baboon Prophylaxis Model

As SYN-005 was shown to be prophylactic in mice and therapeutic in baboons, SYN-005, or its 1B7 component, is being considered for prophylactic use in high-risk areas of the developing world. Injection at birth is expected to protect infants during the first few months of life when the risk of fatality from pertussis is greatest.

In October 2015, the Gates Foundation awarded a grant to the University of Texas at Austin to generate preclinical proof-of-concept data to test the hypothesis that antibody administration at birth may also have a role in the prevention of pertussis. This study to evaluate the potential of Synthetic Biologics' monoclonal antibody, 1B7, to prevent pertussis in the baboon model has initiated.

Safety Profile

No signs of drug-related toxicity were noted in any of the mice or baboons that received SYN-005 in the efficacy studies. The two monoclonal antibodies that comprise SYN-005 are specific for epitopes on pertussis toxin and are not expected to cross react with any human epitopes. Tissue cross reactivity studies and formal toxicology studies will commence with the availability of GMP product in a clinical formulation.

Proposed Indications

Proposed therapeutic indication:
SYN-005 is in development for the treatment of suspected pertussis in newborns and infants up to one year old, to be administered in conjunction with the standard of care antimicrobial therapy

Potential for prophylaxis:
SYN-005 or its individual 1B7 component may be evaluated for prophylaxis of pertussis among newborns whose mothers were unvaccinated in areas with high endemic risk

Proposed Endpoints

Composite endpoints under consideration for the therapeutic indication include

- Reduction in mortality
- Reduction of signs and symptoms
- Normalization of WBC
- Reduction of the neonatal intensive-care unit (NICU)/PICU stay

Proposed Clinical Development Plan

The clinical development plan for this program is intended to focus on at least two global regions where pertussis infection is endemic. Program objectives are to establish dosing regimens that achieve the treatment endpoints (above) and diminish the incidence of clinical pertussis for the prophylactic indication.

Study type	Design	Objective	Regions/Countries
Phase 1 safety Normal adults	Open label	Evaluate pharmacokinetics, safety and tolerability	Americas
Phase 2b Infected infants	Randomized, double blind, controlled, multicenter study Dose ranging	Measure serum levels of SYN-005 over time with the WHO standard as reference Proof-of-principle Evaluate reduction in mortality, decrease in WBC, and prevention of pneumonia	Global (Americas plus other regions)
Phase 3 confirmatory study - Treatment	Randomized, double blind, controlled, multicenter study	Confirmatory One dose level	Global
Phase 3 confirmatory study - Prophylaxis	Randomized, double blind, controlled, multicenter study	Confirmatory One dose level	Global, in geographic areas where pertussis is endemic

Conclusions

- Animal studies support SYN-005 clinical application
- Treatment with SYN-005 is anticipated to shorten hospital and ICU stays for infected infants, diminish the short-term morbidity and long-term sequelae, and prevent fatalities
- Prevention with SYN-005 or its individual 1B7 component is anticipated to protect high-risk newborns from infection during the first few months of life, when the risk of fatality is greatest

SYN-005 has the potential to become the first agent designed to treat and prevent pertussis by neutralizing pertussis toxin

The SYN-005 program is being performed in collaboration with Intrexon Corporation and is based on science, technology, intellectual property, and data developed in collaboration with the University of Texas at Austin's Cockrell School of Engineering, in the laboratory of Assistant Professor, Jennifer Maynard, Ph.D.