Depression Severity and Efficacy Outcomes: Post Hoc Analyses from a Phase 3 Trial of the Novel NMDAR Antagonist Antidepressant Candidate Esmethadone for the Adjunctive Treatment for MDD with Inadequate Response to Standard Antidepressants

Maurizio Fava, MD[1]; Luca Pani, MD[2]; Sara De Martin, PhD[3]; Cornelia Kröger PhD[4], PhD; Paggard Champasa, PhD[4]; Clotilde Guidetti, MD[5]; Stefano Comai, PhD[6]; Andrea Mattarei, PhD[3]; Franco Folli, MD[7]; Sergio Traversa, PharmD[4]; Charles W. Gorodetzky, MD, PhD[8]; Frank J. Vocci, PhD[9]; Frank L. Sapienza, MS[10]; Thomas R. Kosten, MD[11]; Charles E. Inturrisi[4]; David Bushnell, MS[12]; Marin Kappler, PhD[12]; Paolo L. Manfredi, MD[4]; Marco Pappagallo, MD[4]

1 Department of Psychiatry, Massachusetts General Hospital, Boston, MA; 2 Relmada Therapeutics, Inc., Coral Gables, FL; Department of Psychiatry and Behavioral Sciences, School of Medicine, University of Miami; Department of Biomedical, Metabolic and Neural Sciences, University of Modena and Reggio Emilia, Italy; 3 Department of Pharmaceutical and Pharmacological Sciences, University of Padua, Italy ; 4 Relmada Therapeutics, Inc., Coral Gables, FL; 5 Child Neuropsychiatry Unit, Department of Neuroscience, IRCCS Bambino Gesù Pediatric Hospital, Rome Italy; Department of Psychiatry, Massachusetts General Hospital, Boston, MA; 6 Department of Pharmaceutical and Pharmacological Sciences, and Department of Biomedical Sciences, University of Padua, Italy; Department of Psychiatry, McGill University, Montreal, Quebec, Canada; 7 Department of Health Sciences, University of Milan, Italy; 8 Consultant in Pharmaceutical Medicine, Kansas City, MO; 9 Friends Research Institute, Baltimore, MD; 10 The Drug and Chemical Advisory Group LLC, Fairfax, VA; 11 Baylor College of Medicine, MD Anderson Cancer Center, University of Houston, TX; 12 Cytel, Inc., Waltham, MA

INTRODUCTION

- Approximately 50% to 60% of patients with major depressive disorder (MDD) do not obtain an adequate response following their first antidepressant treatment[1]
- Montgomery-Åsberg Depression Rating Scale (MADRS) total score ≥35 is indicative of severe depression [2-5]
- Esmethadone (REL-1017)) is a novel N-methyl-D-aspartate (NMDA) receptor uncompetitive antagonist for the adjunctive treatment of MDD in patients with inadequate response
- In a recently completed Phase 3 study (NCT04688164), esmethadone did not meet the primary efficacy endpoint at Day 28 for mean change from baseline in MADRS. Post-hoc analyses in the subgroup of patients with severe depression (baseline MADRS ≥35) showed efficacy [6]
- Due to biopsychosocial factors, introducing a MADRS score of ≥35 as an eligibility criterion carries the risk of creating clusters of artificially inflated scores that may interfere with informative results [7-10]
- Sensitivity analyses may enhance the reliability of post-hoc primary analyses by showing that the results are consistent across multiple severity cut off points.

AIM

- Sensitivity analyses were conducted to test the validity of post hoc efficacy results of adjunctive esmethadone in the subgroup of patients with severe depression

METHODS

Study Design

Phase 3, double-blind, randomized, placebo-controlled trial of oral once-daily adjunctive esmethadone in adult outpatients with MDD and inadequate response to 1 to 3 antidepressants

Patients 18 to 65 years old were randomly assigned to receive esmethadone (75 mg loading dose on Day 1 and then 25 mg/day thereafter) or placebo for 28 days

Endpoints

- The primary efficacy endpoint was the absolute change from baseline to Day 28 in the MADRS total score
- MADRS total score was analyzed with a mixed model for repeated measures (MMRM) and using mean difference (MD)

Populations

- Prespecified populations for efficacy analyses included:
 - Intent-to-treat (ITT) population: all randomized patients, irrespective of protocol deviations (PDs) or discontinuation
 - Per-protocol (PP) population: patients who completed treatment with no major PDs impacting efficacy assessments
- Post-hoc analyses were performed in:
 - Subgroup of patients with a MADRS score ≥35 at baseline

Sensitivity Analyses

- Sensitivity analyses were performed at different severity cut off points (31, 32, 33, 34, 35, 36, 37) for the ITT and PP populations.
- Change from baseline was modeled individually in each treatment arm by:
 1) ANCOVA model allowing non-parallel slopes of MADRS baseline value
 2) Smoothing spline regression model. Smoothing spline regression is a non-parametric regression technique used to fit a smooth curve through a set of data points

REFERENCES

1. Fava M, Davidson KG. Definition and epidemiology of treatment-resistant depression. *Psychiatr Clin North Am*. 1996;19:179-200.
2. Montgomery SA, Asberg M. A new depression scale designed to be sensitive to change. Br J Psychiatry. 1979 Apr;134:382-9.
3. Müller MJ, Szegedi A, Wetzel H, Benkert O. Moderate and severe depression. Gradations for the Montgomery-Asberg Depression Rating Scale. J Affect Disord. 2000 Nov;60(2):137-40.
4. Herrmann N, Black SE, Lawrence J, Szekely C, Szalai JP. The Sunnybrook Stroke Study: a prospective study of depressive symptoms and functional outcome. Stroke. 1998 Mar;29(3):618-24.
5. American Psychiatric Association. (2013). Diagnostic and statistical.
6. Fava M, Stahl SM, Pani L et al. Efficacy and Safety of Esmethadone (REL-1017) in Patients With Major Depressive Disorder and Inadequate Response to Standard Antidepressants: A Phase 3 Randomized Controlled Trial. J Clin Psychiatry. 2024 Jun 17;85(3):24m15265.
7. Agid O, Siu CO, Potkin SG et al. Meta-regression analysis of placebo response in antipsychotic trials, 1970-2010. Am J Psychiatry 2013;170:1335-44.
8. Correll CU, Solmi M, Cortese S, et al. The future of psychopharmacology: a critical appraisal of ongoing phase 2/3 trials, and of some current trends aiming to de-risk trial programmes of novel agents. World Psychiatry. 2023;22(1):48-74.
9. Khan A, Schwartz K, Kolts RL, Ridgway D, Lineberry C. Relationship between depression severity entry criteria and antidepressant clinical trial outcomes. Biol Psychiatry. 2007;62(1):65-71.
10. Landin R, DeBrota DJ, DeVries TA, Potter WZ, Demitrack MA. The impact of restrictive entry criterion during the placebo lead-in period. Biometrics. 2000; 56:271–278.

DISCLOSURES

Drs. Pappagallo and Manfredi contributed equally. This work was funded by Relmada Therapeutics, Inc. Drs. De Martin, Guidetti, Mattarei, and Comai are employed by or have received compensation from companies or institutions that received funding from Relmada Therapeutics, Inc. Dr. Fava is a consultant to Relmada on behalf of Massachusetts General Hospital and did not receive any personal compensation. Drs. Pani, Gorodetzky, Vocci, Sapienza, Kosten, Folli, Kröger, Champasa, Manfredi, Pappagallo, and Inturrisi have received consultant fees from Relmada Therapeutics, Inc. Dr.Traversa is an employee of Relmada Therapeutics, Inc. Drs. De Martin, Mattarei and Comai have received grant support from MGGM LLC and consultant fees from Neuroarbor LLC. Drs. Guidetti has received consultant fees from MGGM LLC. Drs. Inturrisi and Manfredi are co-inventors of technology related to esmethadone.

RESULTS

Table 1. MMRM Analyses for Intent-To-Treat, Per Protocol, and Post-hoc Severe Depression Population for Mean Change from Baseline to Day 28 (Primary Endpoint)

	Esmethadone	Placebo	LS Mean Difference (esmethadone – placebo)
Intent to Treat	N=113	N=114	
Baseline, mean (SD)	34.7 (5.2)	35.3 (4.3)	
LS Mean (SE)	-15.10 (1.05)	-13.37 (1.09)	-1.74 (1.52)
95% CI	-17.18, -13.02	-15.52, -11.22	-4.73, 1.26
p-value			0.255
Effect size			-0.16
Per Protocol	N=101	N=97	
Baseline, mean (SD)	34.6 (5.3)	35.1 (4.4)	
LS Mean (SE)	-15.63 (1.06)	-12.69 (1.10)	-2.94 (1.53)
95% CI	-17.73, -13.54	-14.87, -10.51	-5.96, 0.08
p-value			0.057
Effect size			-0.28
MADRS ≥35	N=51	N=61	
Baseline, mean (SD)	39.4 (3.3)	38.3 (2.9)	
LS Mean (SE)	-17.87 (1.70)	-11.83 (1.58)	-6.04 (2.33)
95% CI	-21.24, -14.50	-14.97, -8.70	-10.65, -1.42
p-value			0.011
Effect size			-0.51

Effect Size = Cohen's D effect size

Table 2. Mean Difference (MD) Analyses for Intent-To-Treat, Per Protocol, and Post-hoc Severe Depression Population for Mean Change from Baseline to Day 28

	Esmethadone			Placebo			Difference Drug Minus Placebo			
	N (missing*)	Mean	SD	N (missing*)	Mean	SD	Mean	SD	p-value	Effect Size
ITT (n=227)	103 (10)	-15.1	11.3	88 (26)	-12.9	10.4	-2.3	10.9	0.154	-0.21
PP (n=198)	97 (4)	-15.6	11.2	84 (13)	-12.5	9.9	-3.1	10.6	0.051	-0.29
MADRS ≥35 (n=112)	48 (3)	-18.5	13.3	51 (10)	-11.7	10.9	-6.9	12.1	0.006	-0.57

Table 3: Day 28 Response and Remission Rates for Intent-To-Treat, Per Protocol, and Post-hoc Severe Depression Population

	Esmethadone	Placebo	Rate Difference	p-value	Odds ratio	95% Confidence Interval
Response						
ITT (n=227)	39.8%	27.2%	12.6%	0.044	1.77	0.98, 3.23
PP (n=198)	42.6%	29.9%	12.7%	0.064	1.74	0.93, 3.27
MADRS ≥35 (n=112)	43.1%	21.3%	21.8%	0.013	2.80	1.14, 7.00
Remission						
ITT (n=227)	22.1%	13.2%	9.0%	0.076	1.88	0.88, 4.08
PP (n=198)	23.8%	13.4%	10.4%	0.062	2.01	0.91, 4.61
MADRS ≥35 (n=112)	27.5%	11.5%	16.0%	0.031	2.92	0.98, 9.33

Patients with missing assessments were considered non-responders at that time point. P-values are based on a Chi-square test. Response: ≥50% improvement from baseline MADRS score at Day 28. Remission: MADRS score ≤10 at Day 28.

Table 4. MMRM and Mean Difference (MD) Analyses for Intent-To-Treat (ITT) and Per Protocol (PP) Populations for Mean Change from Baseline to Day 28

MADRS	Population	Number	LS Mean (SE)	P-value	Effect Size	MD (SD)	P-value	Effect Size
≥31	ITT	181	-2.75 (1.77)	0.1228	-0.24	-3.5 (11.5)	0.0587	-0.31
≥31	PP	157	-3.99 (1.78)	0.0266	-0.36	-4.5 (11.1)	0.0165	-0.40
≥32	ITT	169	-3.01 (1.87)	0.1092	-0.26	-3.9 (11.6)	0.0467	-0.33
≥32	PP	145	-4.39 (1.89)	0.0214	-0.39	-4.9 (11.3)	0.0119	-0.44
≥33	ITT	156	-4.20 (1.94)	0.0318	-0.36	-4.8 (11.6)	0.0188	-0.41
≥33	PP	133	-5.78 (1.94)	0.0035	-0.52	-6.0 (11.2)	0.0034	-0.53
≥34	ITT	135	-4.57 (2.14)	0.0344	-0.38	-5.3 (12.0)	0.0184	-0.44
≥34	PP	117	-5.98 (2.16)	0.0065	-0.52	-6.3 (11.6)	0.0049	-0.55
≥35	ITT	112	-6.04 (2.33)	0.0109	-0.51	-6.9 (12.1)	0.0059	-0.57
≥35	PP	98	-7.25 (2.32)	0.0024	-0.64	-7.9 (11.6)	0.0015	-0.68
≥36	ITT	96	-5.92 (2.51)	0.0208	-0.50	-6.7 (12.0)	0.0123	-0.56
≥36	PP	83	-7.39 (2.50)	0.0040	-0.66	-7.8 (11.4)	0.0029	-0.69
≥37	ITT	75	-6.68 (2.92)	0.0254	-0.55	-7.9 (12.3)	0.0117	-0.65
≥37	PP	63	-7.92 (2.91)	0.0086	-0.69	-8.8 (11.8)	0.0051	-0.75

Figure 3: Relation of MADRS score at baseline and change from baseline at Day 28 by treatment arm – ANCOVA

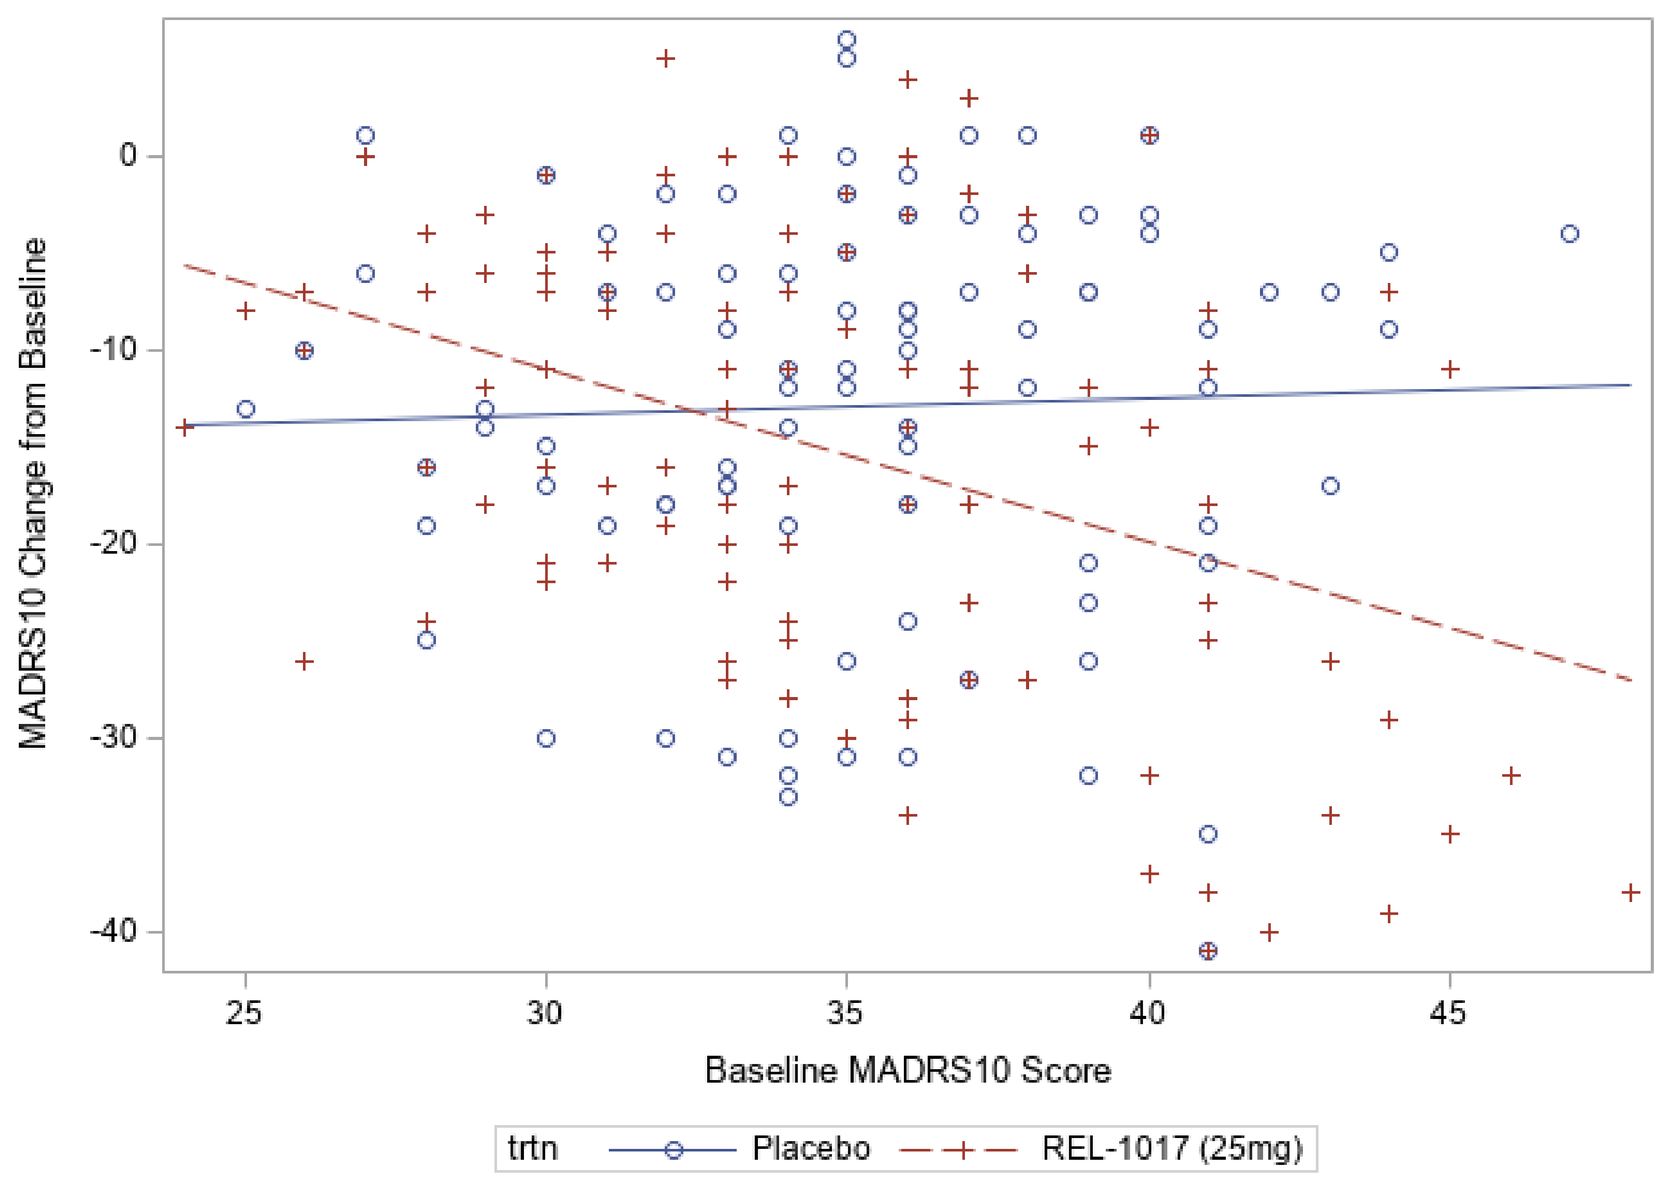
MADRS10 Change from Baseline
Baseline MADRS10 Score
trtn
Placebo
REL-1017 (25mg)

Figure 4: Relation of MADRS score at baseline and change from baseline at Dat 28 by treatment arm - Smoothing spline model

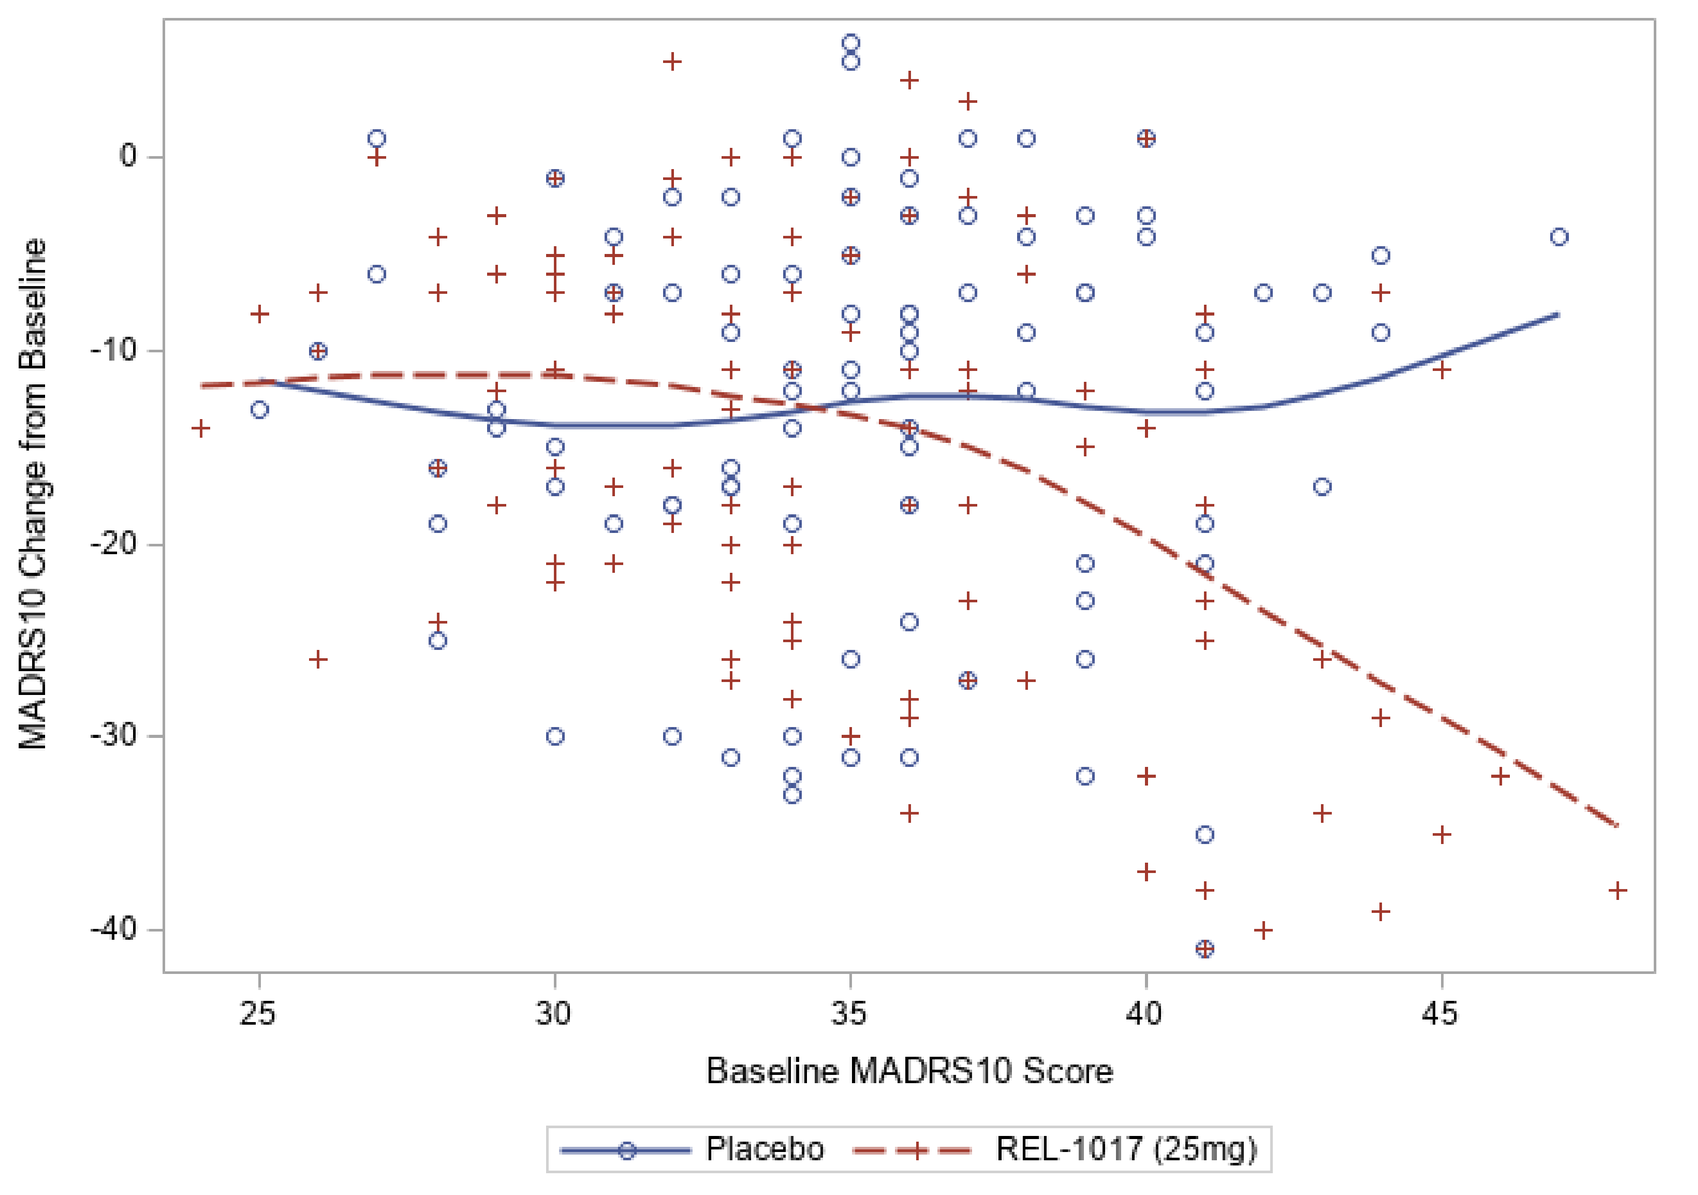
MADRS10 Change from Baseline
Baseline MADRS10 Score
Placebo
REL-1017 (25mg)

Figure 1: Day 28 Response and Remission Rate for Intent-To-Treat, Per Protocol, and Post-hoc Severe Depression Population

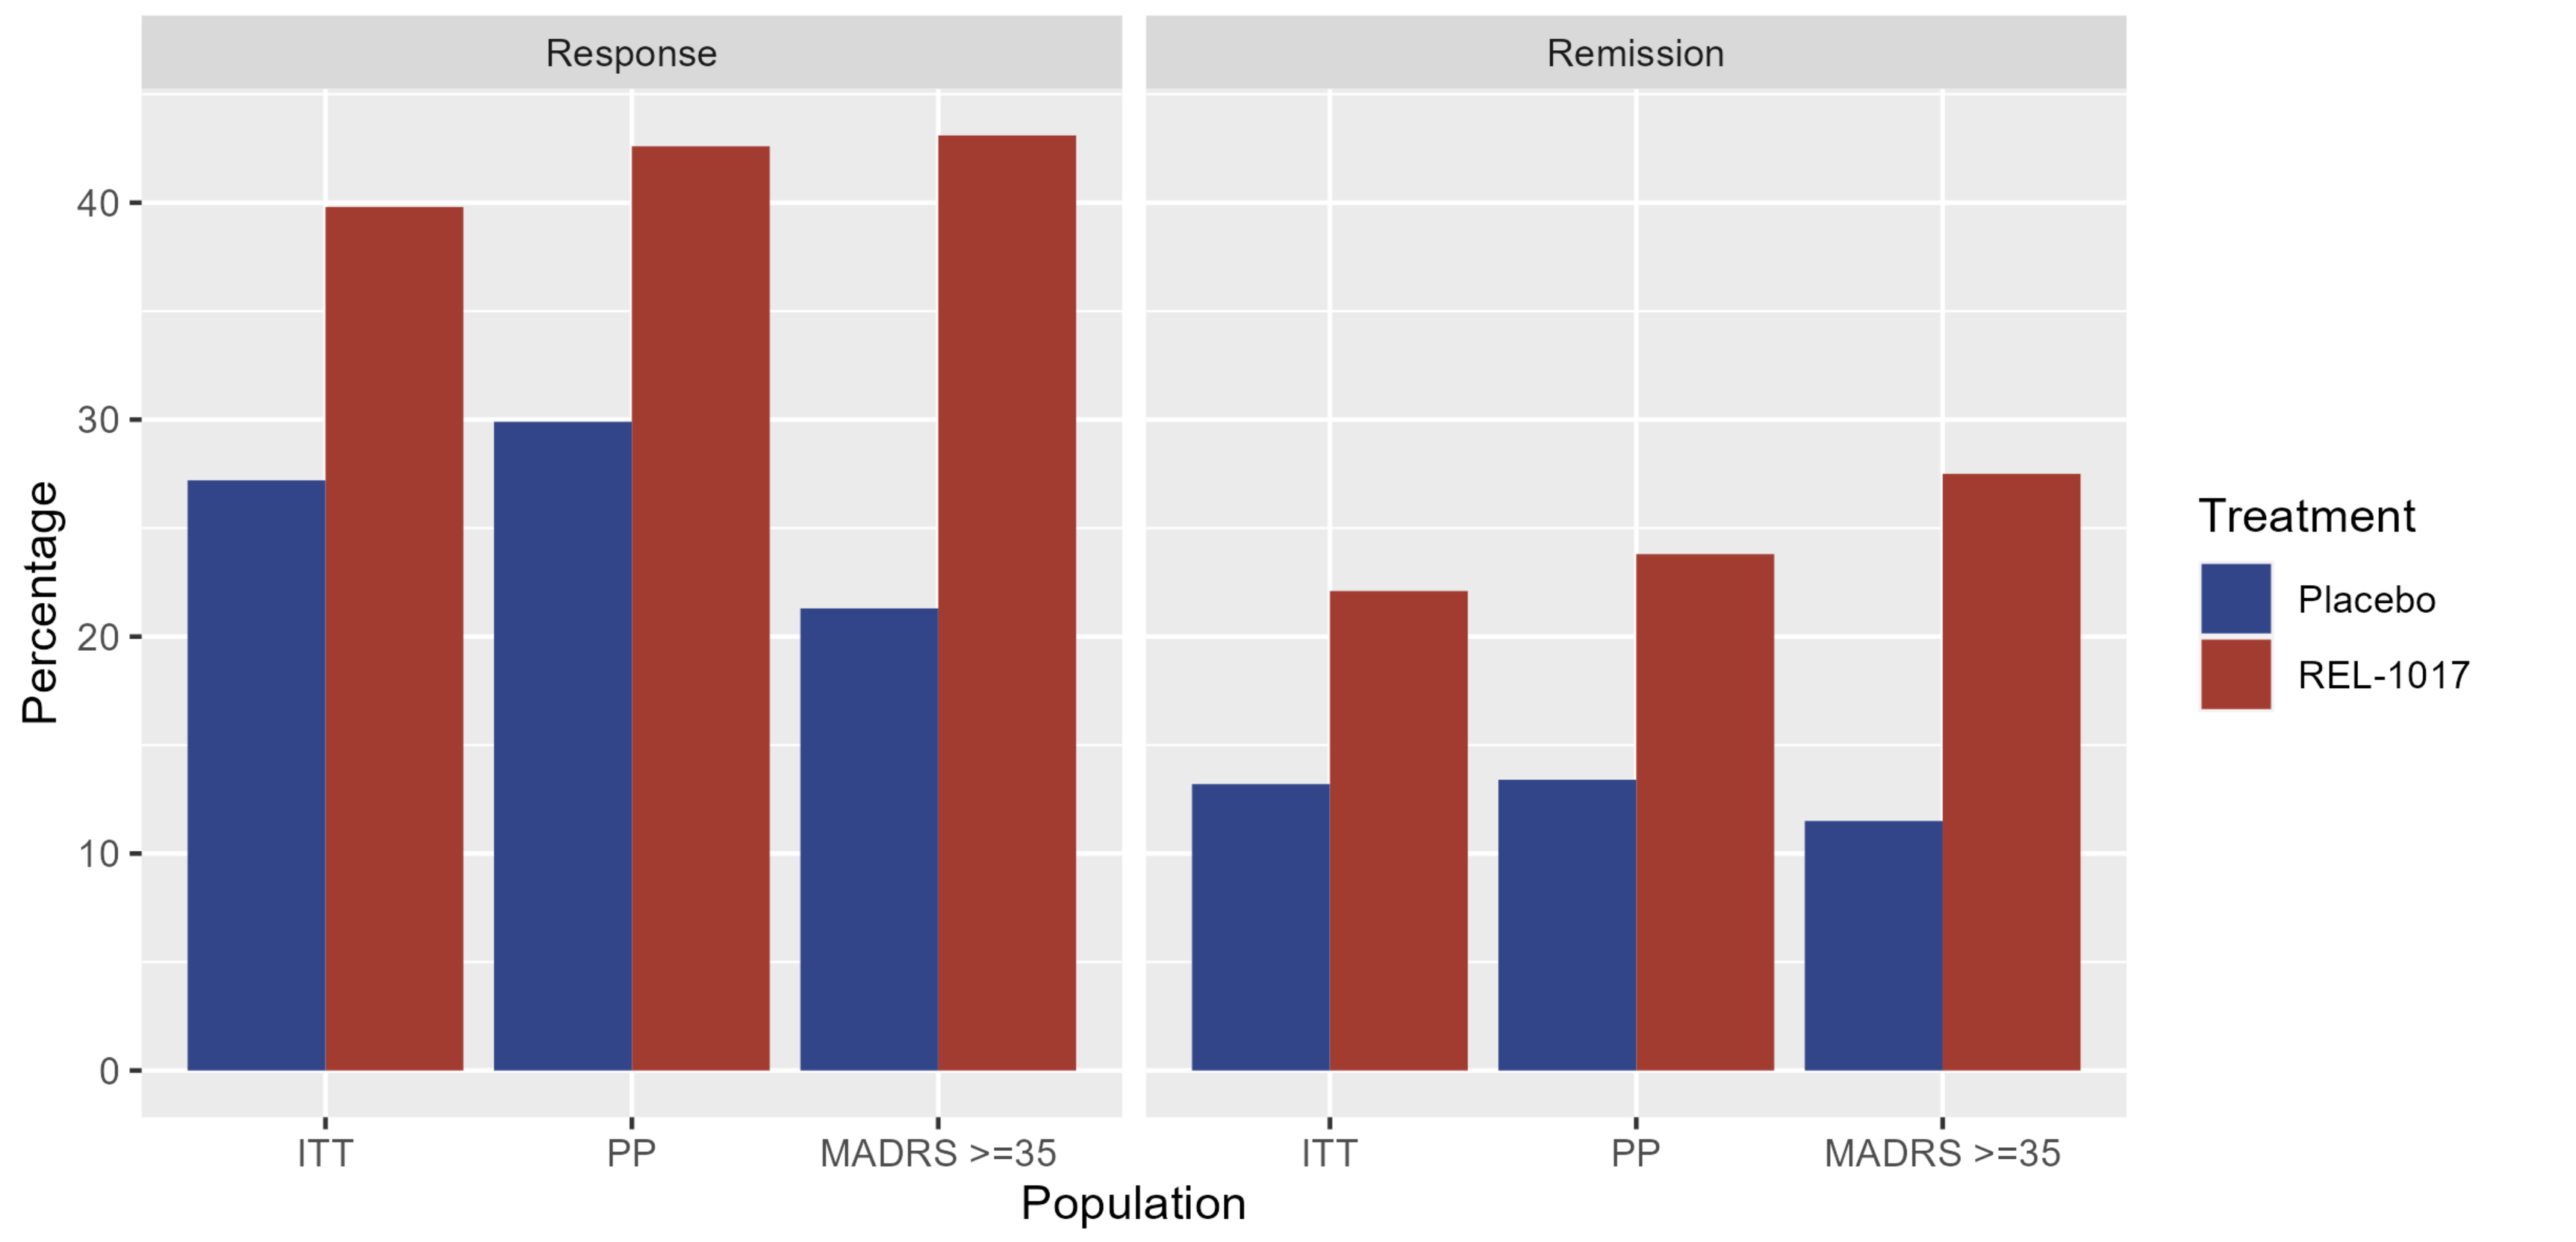
Response
Remission
Percentage
ITT
PP
MADRS >=35
Population
Treatment
Placebo
REL-1017

Response: ≥50% improvement from baseline MADRS score at Day 28; Remission: MADRS score ≤10 at Day 28

Figure 2: Effect size estimated by MMRM and Mean Difference analyses on patient subpopulations with severe depression at baseline ≥ a given cutoff (ITT and PP populations)

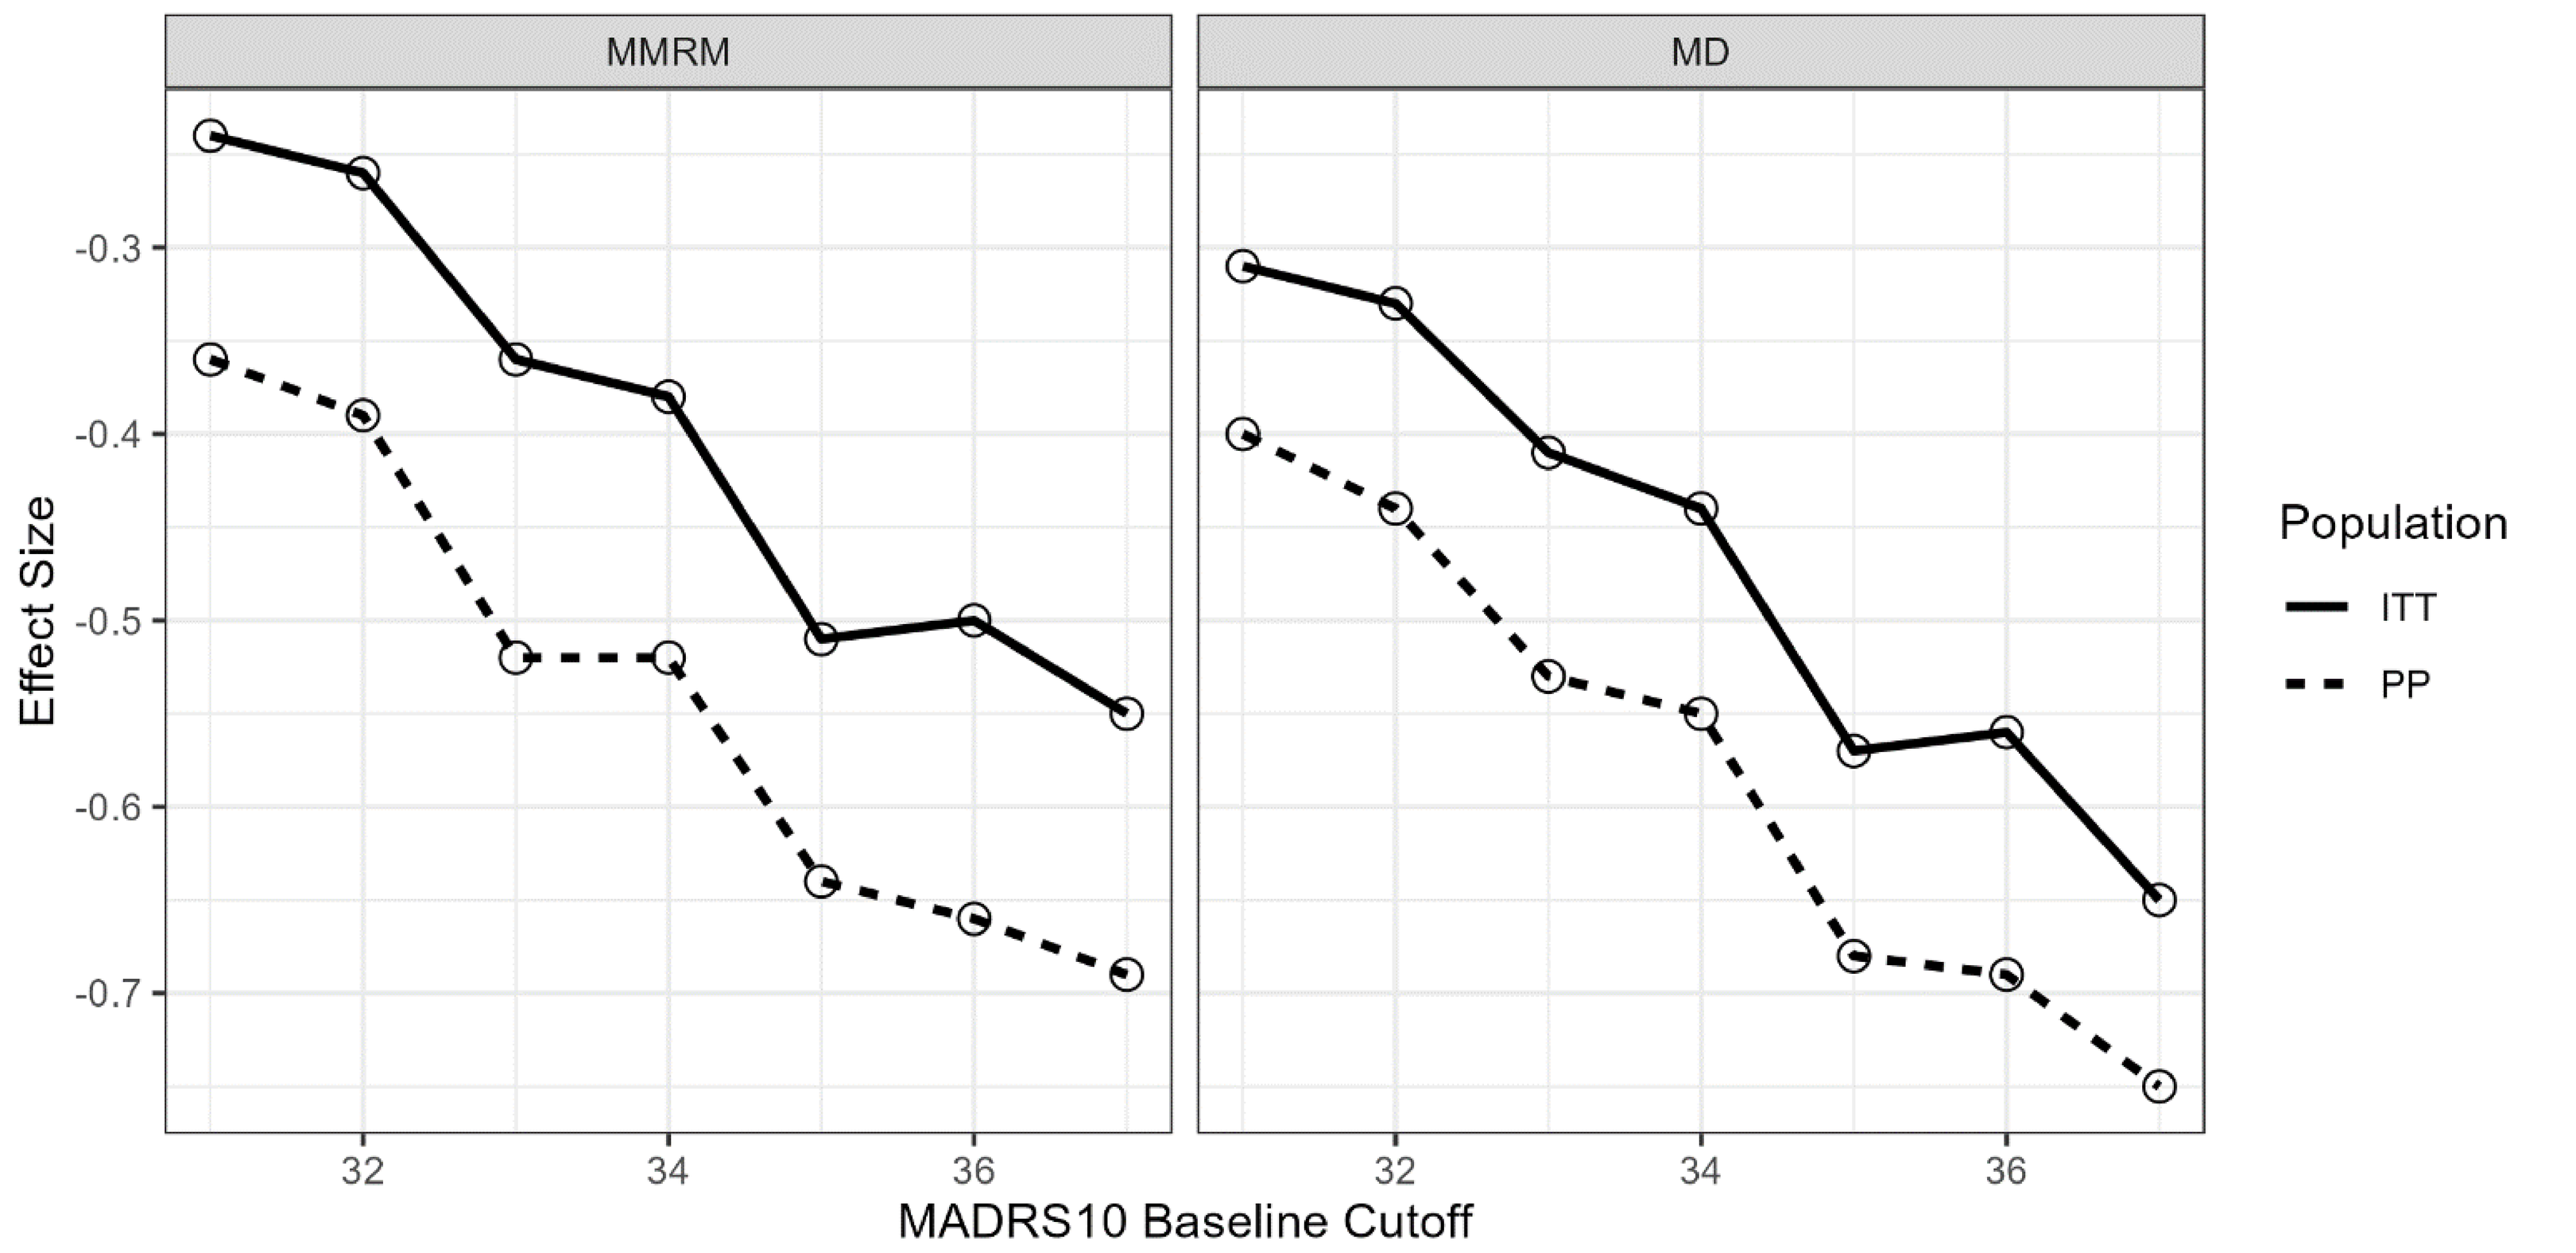
MMRM
MD
Effect Size
MADRS10 Baseline Cutoff
Population
ITT
PP

- Post-hoc analyses in the patient subgroup with severe depression (baseline MADRS ≥35), showed efficacy with robust effect size in the MMRM analyses (Table 1) and in the Mean Difference Analyses (Table 2).
- While the difference in response rate and the difference in remission rate for esmethadone vs. placebo appear meaningful in the ITT and PP populations, the severe depression population showed significant improvement for both parameters (Table 3 and Figure 1).
- Post-hoc analyses carry the risk of Type 1 statistical error. The validity of the results achieved with esmethadone in severe MDD (baseline MADRS ≥35) was tested with analyses at different severity cutoff points (Table 4 and Figure 2).
- In additional post-hoc sensitivity analyses targeting the relation between depression severity at baseline and the treatment effect, we modeled the change from baseline individually in each treatment arm:
- In the ANCOVA model with the esmethadone arm, the regression line starts at around -7 for the lowest baseline MADRS score (~25) and decreases to between -25 and -28 for MADRS baseline >45 (Figure 3). In the placebo arm, the change from baseline regression line is stable with values between -14 to -12 over the whole range of the baseline MADRS scores. Overall, the treatment difference at Day 28 was influenced by baseline MADRS:
 - Treatment difference at baseline MADRS=32 (Q1): +0.4 (placebo better; p = 0.82)
 - Treatment difference at baseline MADRS=35 (median): -2.5 (p = 0.10)
 - Treatment difference at baseline MADRS=38 (Q3): -5.4 (p = 0.003)
- Results with the smoothing spline model were similar to the ANCOVA model for the placebo arm (Figure 4). MADRS change from baseline remained stable at -12 to -14 with a slight increase at higher MADRS baseline scores. In the esmethadone arm, no noticeable difference vs. placebo was seen for baseline MADRS scores <35, but the curves separate for baseline MADRS above 35. MADRS change from baseline decreased from -12 (MADRS baseline = 35) to -30 (MADRS baseline = 45) corresponding to a slope of -1.8.

CONCLUSIONS

- **Baseline depression severity seems to be a relevant factor for treatment response to esmethadone. The target population for the primary endpoint most appropriate to demonstrate esmethadone superiority vs. placebo seems be the subgroup of patients with severe depression baseline (MADRS ≥35).**
- **Efficacy studies of esmethadone and other antidepressant candidates should include patients with moderate depression (e.g., baseline MADRS ≥24). However, when testing esmethadone, patients with severe depression should be prespecified as the target population for the primary endpoint.**

ACNP December 8-11, 2024

Relmada THERAPEUTICS

Corresponding author: Paolo Manfredi, MD Email: pmanfredi@relmada.com