Chronic non-psychedelic psilocybin therapy promotes visual rehabilitation in mice post-occipital stroke

S. NASINI[1], B. BARZON[1], M. CAMBIAGHI[2], S. DE MARTIN[1], A. MATTAREI[1], M. PAPPAGALLO[3], P. MANFREDI[3], ***S. COMAI**[1];
[1]Univ. of Padova, Padova, Italy; [2]Dep. Neurosci., Univ. of Verona, Verona, Italy; [3]Relmada Therapeutics, Inc, Coral Gables, FL

LBA003.142

INTRODUCTION

Stroke is one of the leading causes of mortality and long-term disability worldwide, with profound impact on individuals and healthcare systems. Among the different types of strokes, occipital stroke is particularly debilitating, as it directly affects the brain's visual processing centers, often leading to significant visual and functional deficits *(Kerkhoff, 2000)*. Occipital strokes are usually unilateral, causing damage to one hemisphere of the visual cortex, with severe impairments in visual perception and processing. As a result, patients may experience a substantial decline in their quality of life. Currently, there are no effective treatments to restore visual function following an occipital stroke, highlighting an urgent need for innovative therapeutic approaches. Emerging research has pointed to the potential of psychedelics, such as psylocibin, to induce neuroplasticity and promote neurogenesis *(Ly et al., 2018)*. These mechanisms suggest that psychedelics, known for inducing visual hallucinations, may have a particular affinity for visual pathways. Consequently, we hypothesized that psilocybin may stimulate visual pathway restoration following occipital stroke, thereby potentially opening new avenues for improving patient outcomes and reducing long-term disability.

METHODS

This study investigates the effects of a chronic treatment (45 days) with a low non-psychedelic dose of psilocybin (0.05 mg/Kg) on visual recovery in adult (2-3-month-old) mice subjected to photothrombotic monolateral stroke *(Cambiaghi et al., 2022)* at the visual cortex level. Visual abilities were evaluated after 45 days of psilocybin post- stroke treatment using the Visual Cliff test, the Looming test, and the Cued Morris Water Maze test.

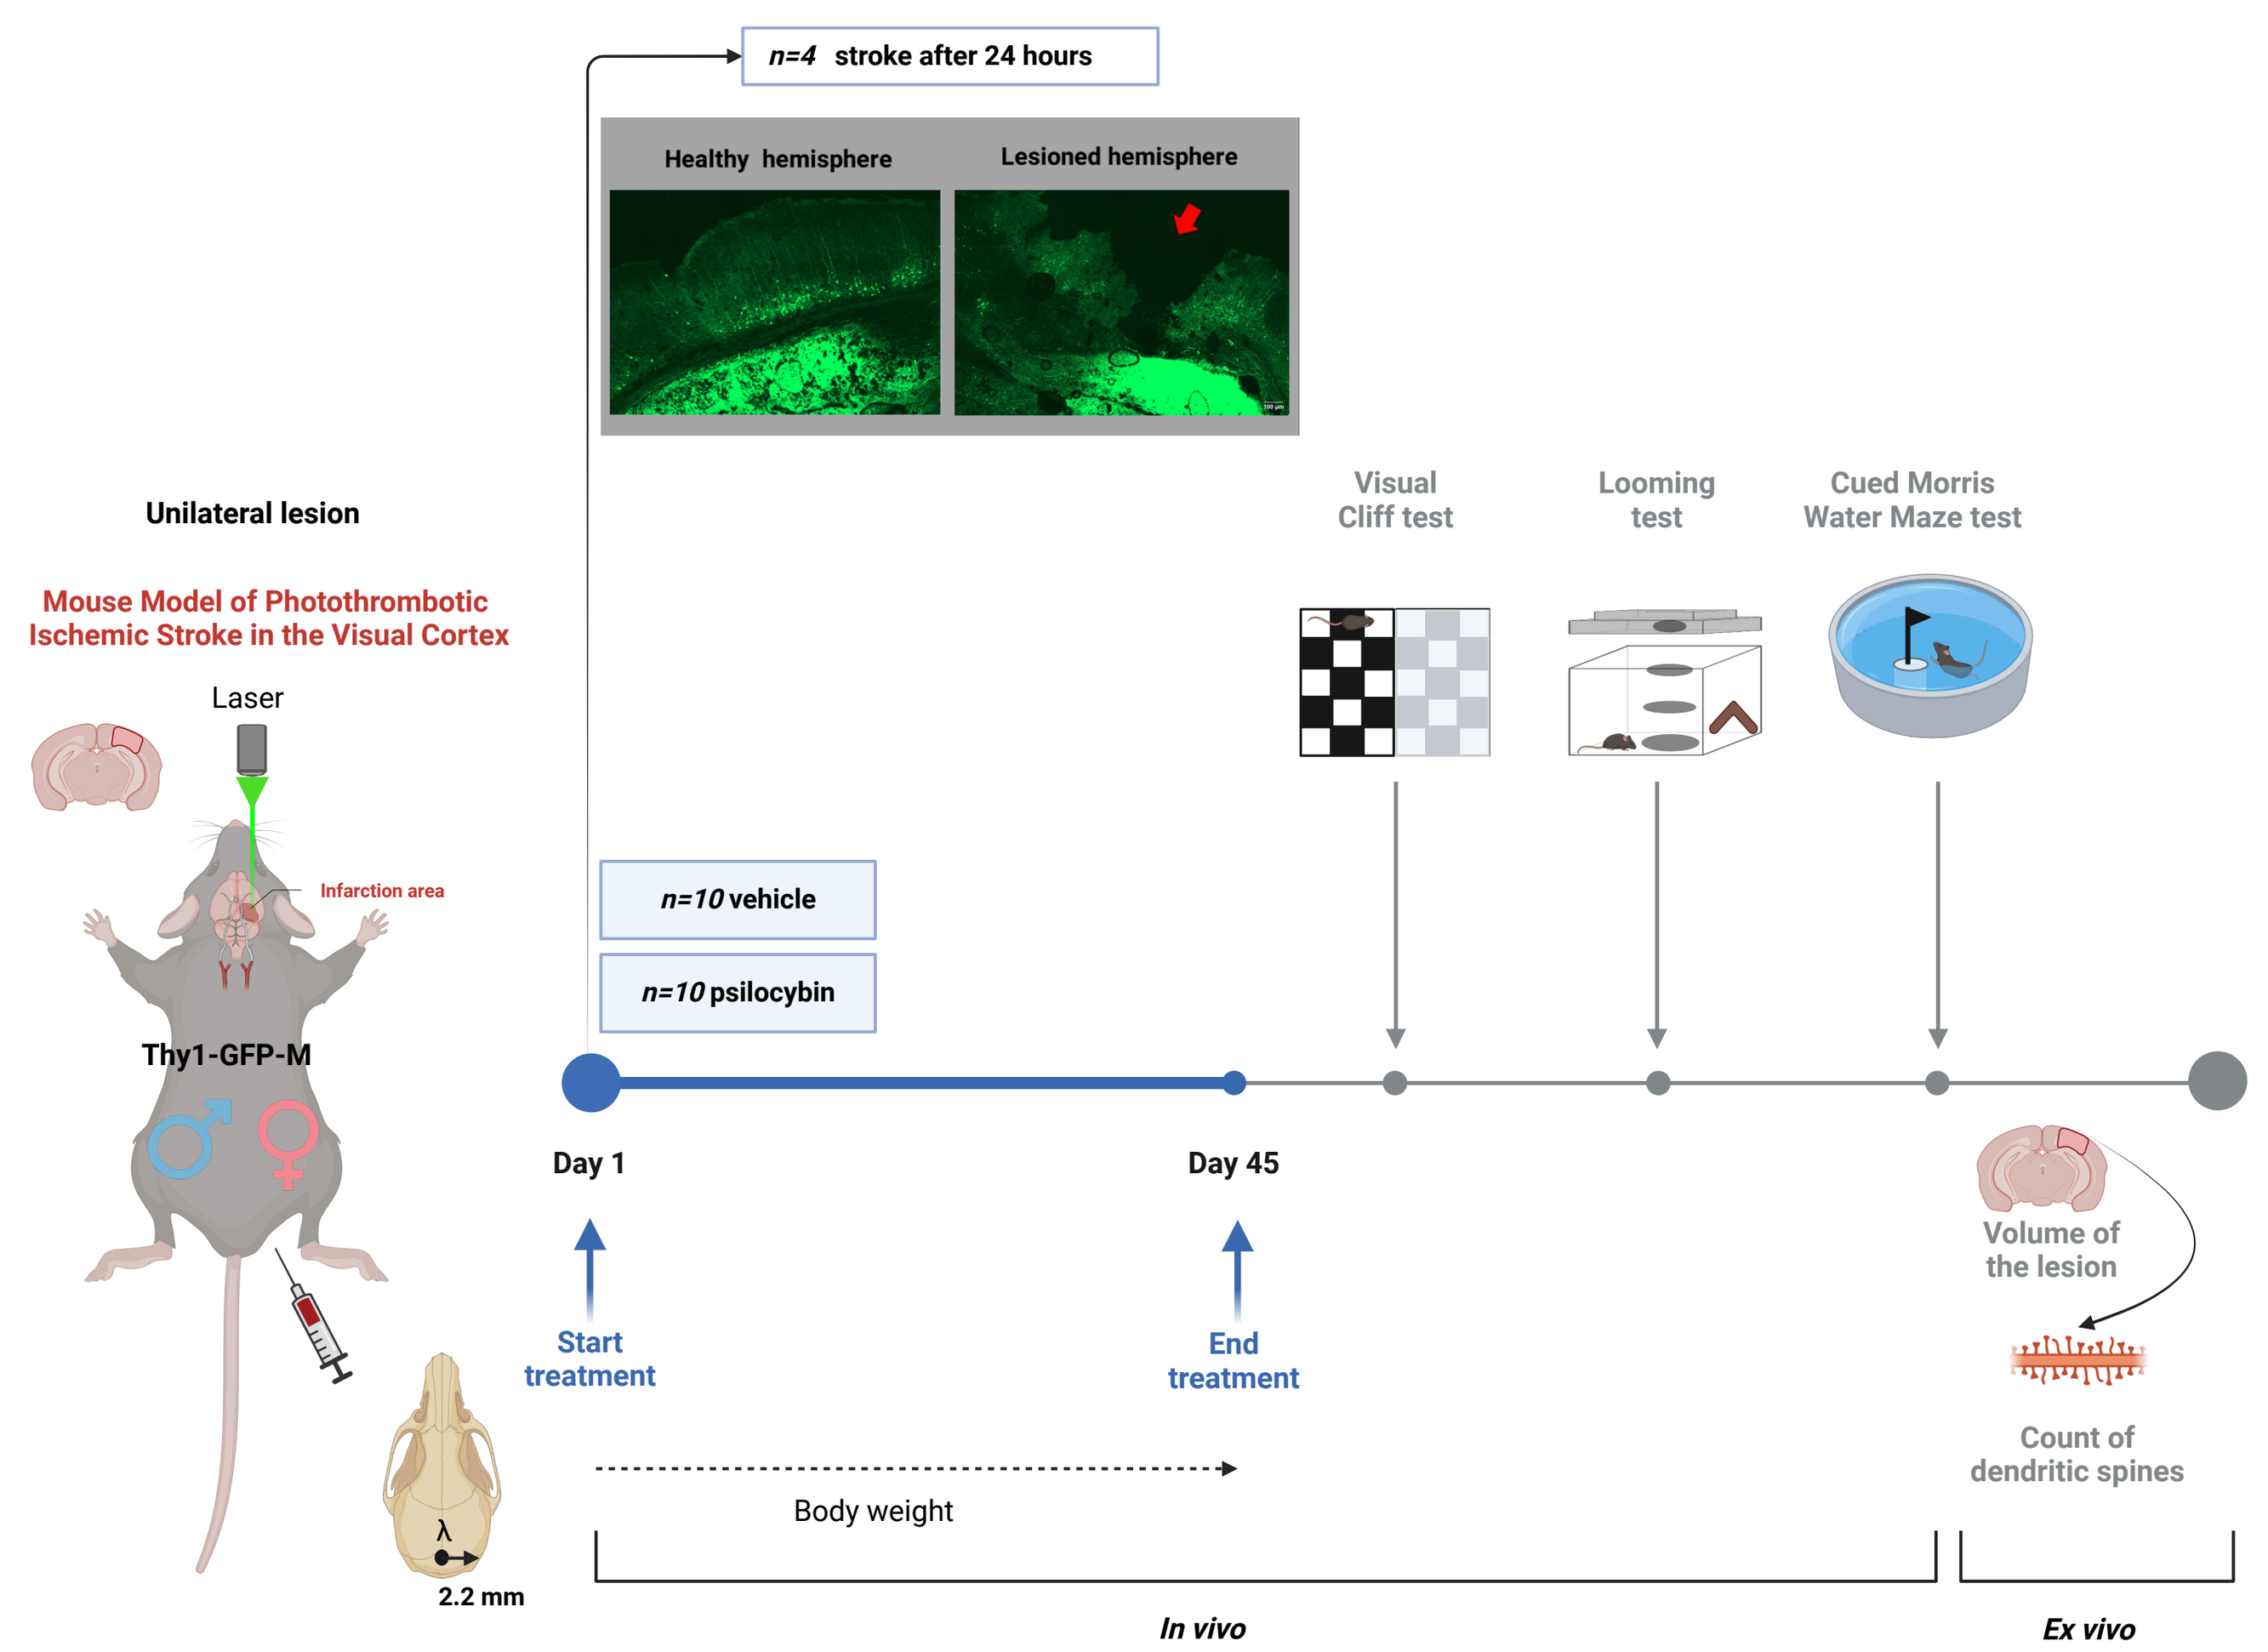
n=4 stroke after 24 hours
Healthy hemisphere
Lesioned hemisphere
Unilateral lesion
Mouse Model of Photothrombotic Ischemic Stroke in the Visual Cortex
Laser
Infarction area
Thy1-GFP-M
2.2 mm
Visual Cliff test
Looming test
Cued Morris Water Maze test
n=10 vehicle
n=10 psilocybin
Day 1
Day 45
Start treatment
End treatment
Volume of the lesion
Count of dendritic spines
Body weight
In vivo
Ex vivo

RESULTS

VISUAL CLIFF TEST

Psilocybin treatment restores speed in the depth zone to control levels

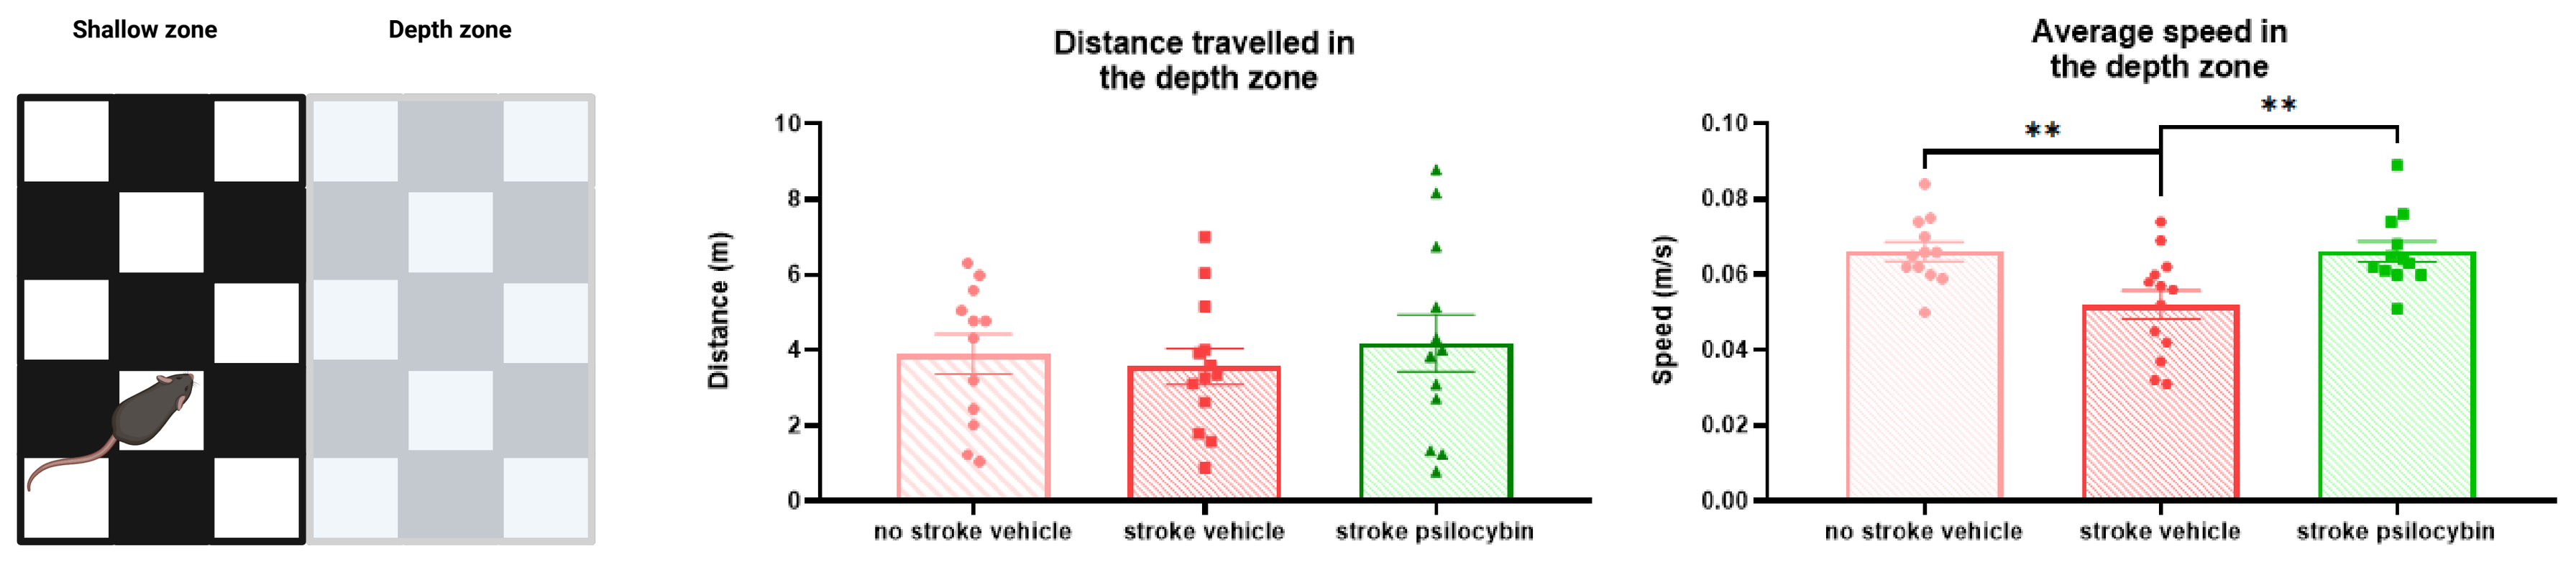
Shallow zone
Depth zone
Distance travelled in the depth zone
Distance (m)
Average speed in the depth zone
Speed (m/s)
no stroke vehicle
stroke vehicle
stroke psilocybin
**

LOOMING VISUAL STIMULI TEST

Psilocybin decreases the time to enter the safe zone to control levels

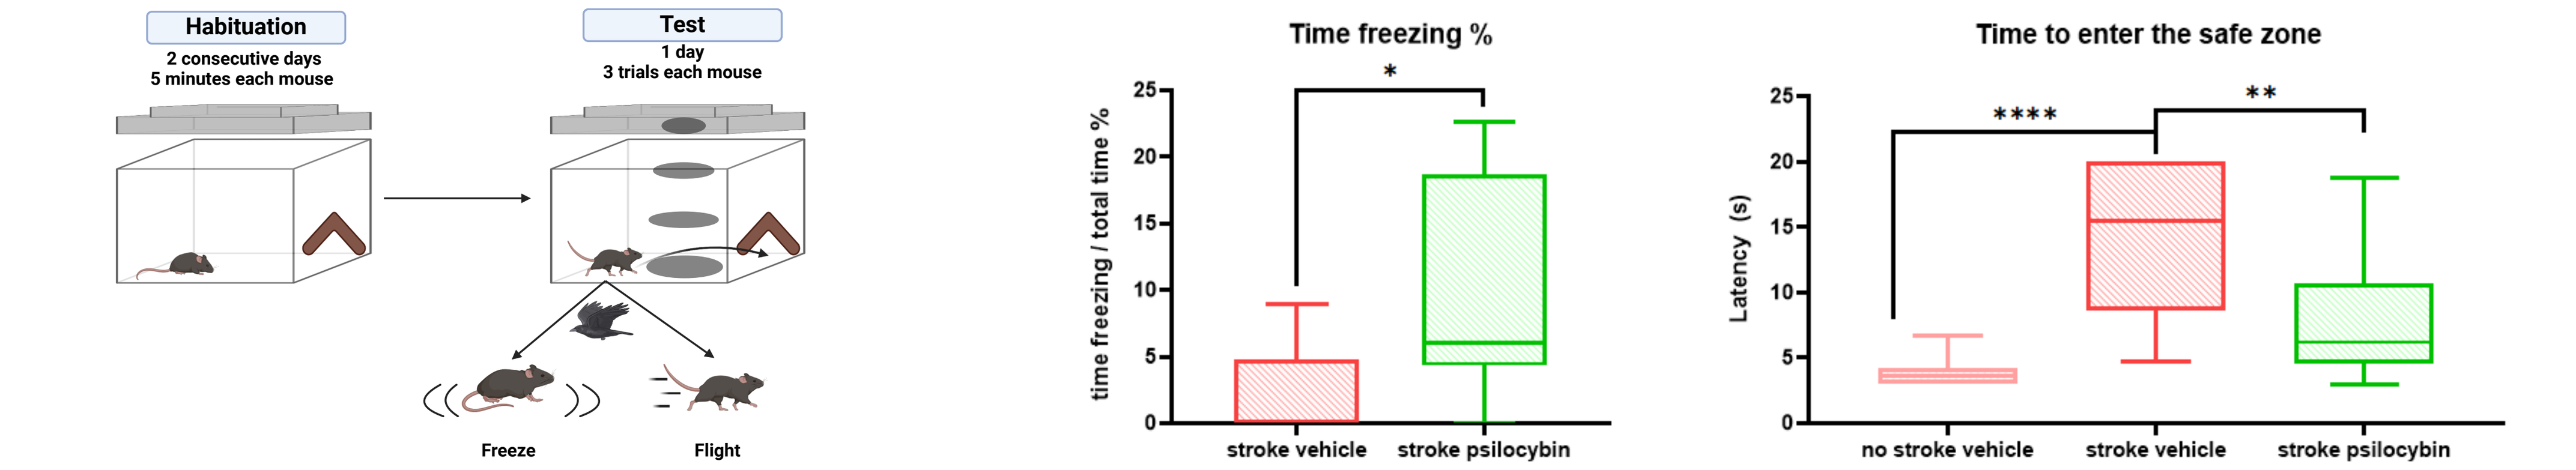
Habituation
2 consecutive days
5 minutes each mouse
Test
1 day
3 trials each mouse
Freeze
Flight
Time freezing %
time freezing / total time %
Time to enter the safe zone
Latency (s)
no stroke vehicle
stroke vehicle
stroke psilocybin

**
*

CUED MORRIS WATER MAZE TEST

Psilocybin-treated mice reach the platform more times than vehicle-treated mice on day 1 of the test

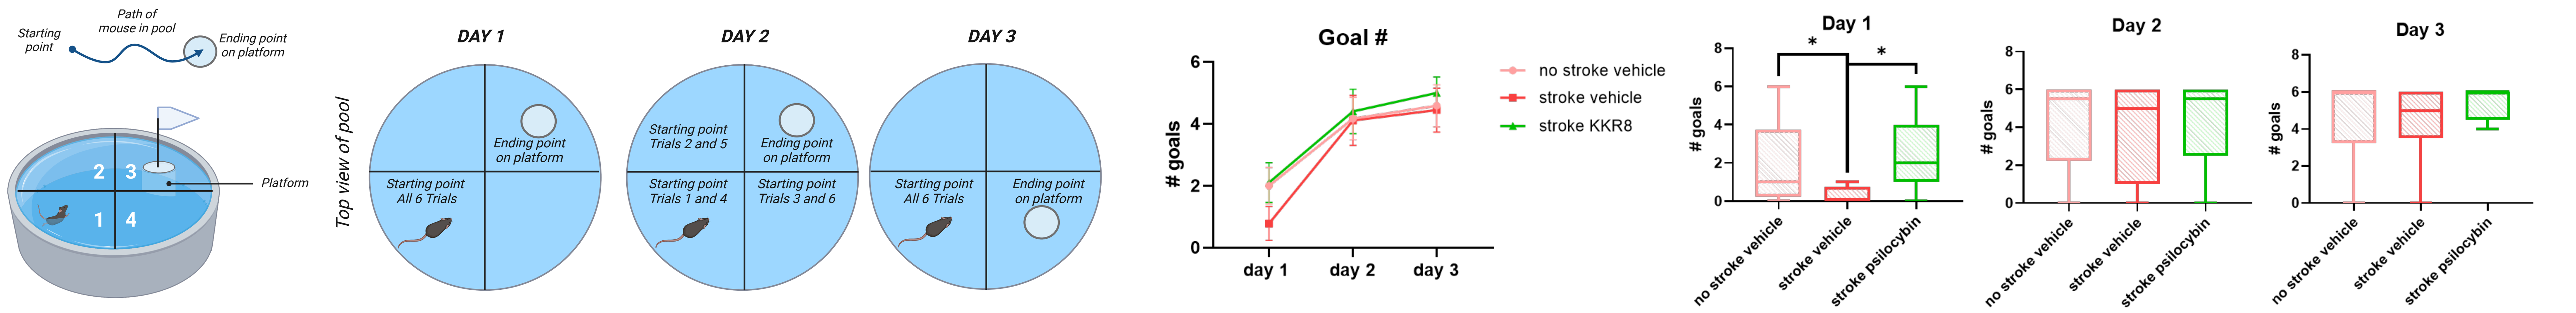
Starting point
Path of mouse in pool
Ending point on platform
Platform
Top view of pool
DAY 1
DAY 2
DAY 3
Starting point All 6 Trials
Starting point Trials 2 and 5
Starting point Trials 1 and 4
Starting point Trials 3 and 6
Goal #
goals
day 1
day 2
day 3
no stroke vehicle
stroke vehicle
stroke KKR8
Day 1
Day 2
Day 3
stroke psilocybin

Volume of the lesion after 45 days of treatment

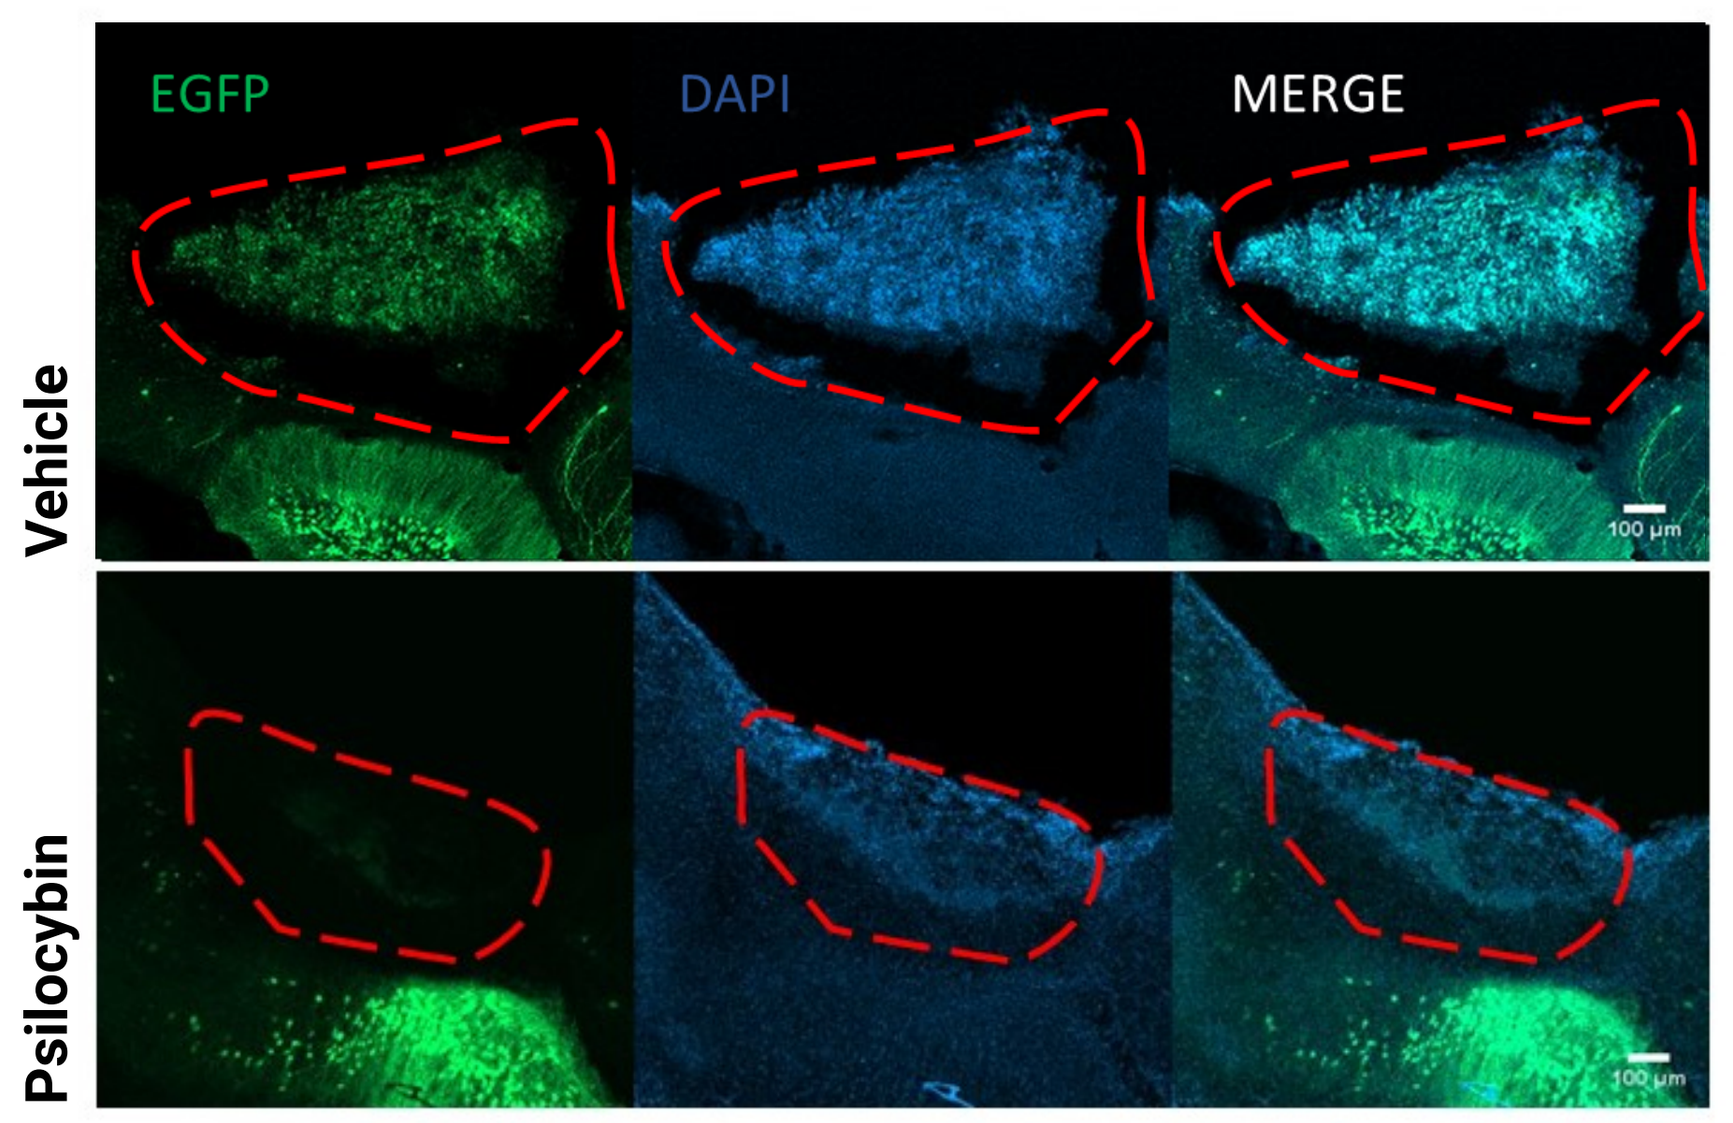
EGFP
DAPI
MERGE
Vehicle
Psilocybin
100 µm

Images were acquired using a ZEISS LSM 800 Confocal Laser Scanning Microscope (Carl Zeiss, Jena, Germany) equipped with a Zeiss 5x zoom 1.0x

After 45 days both vehicle- and psilocybin-treated animals show reduced volume of lesion compared to 24 hours post-stroke.

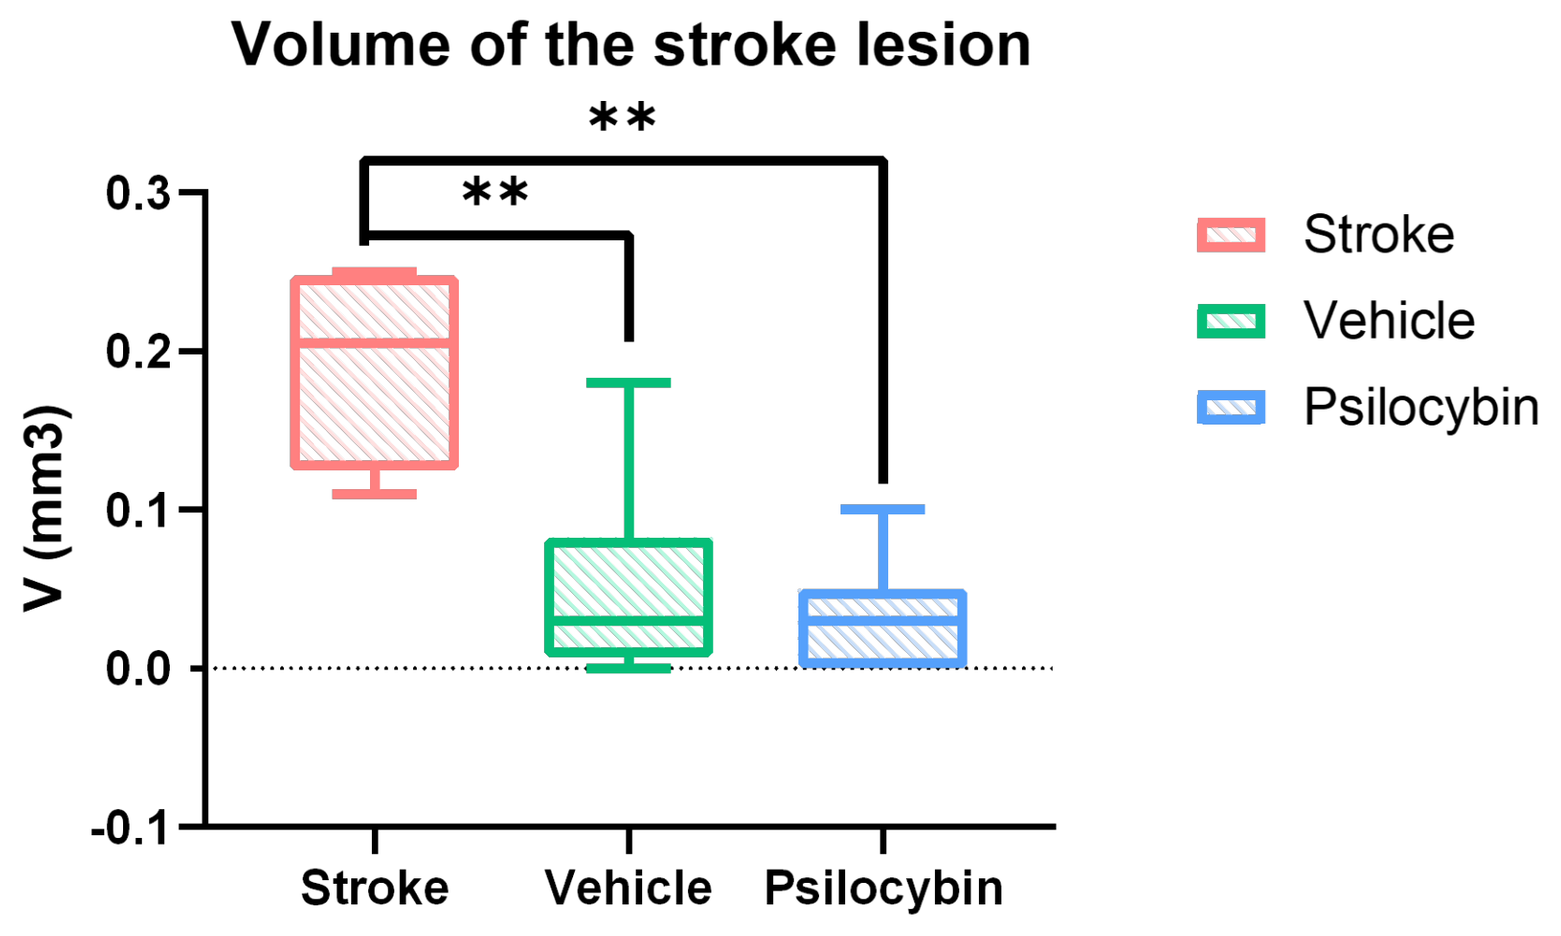
Volume of the stroke lesion
V (mm3)
Stroke
Vehicle
Psilocybin
**

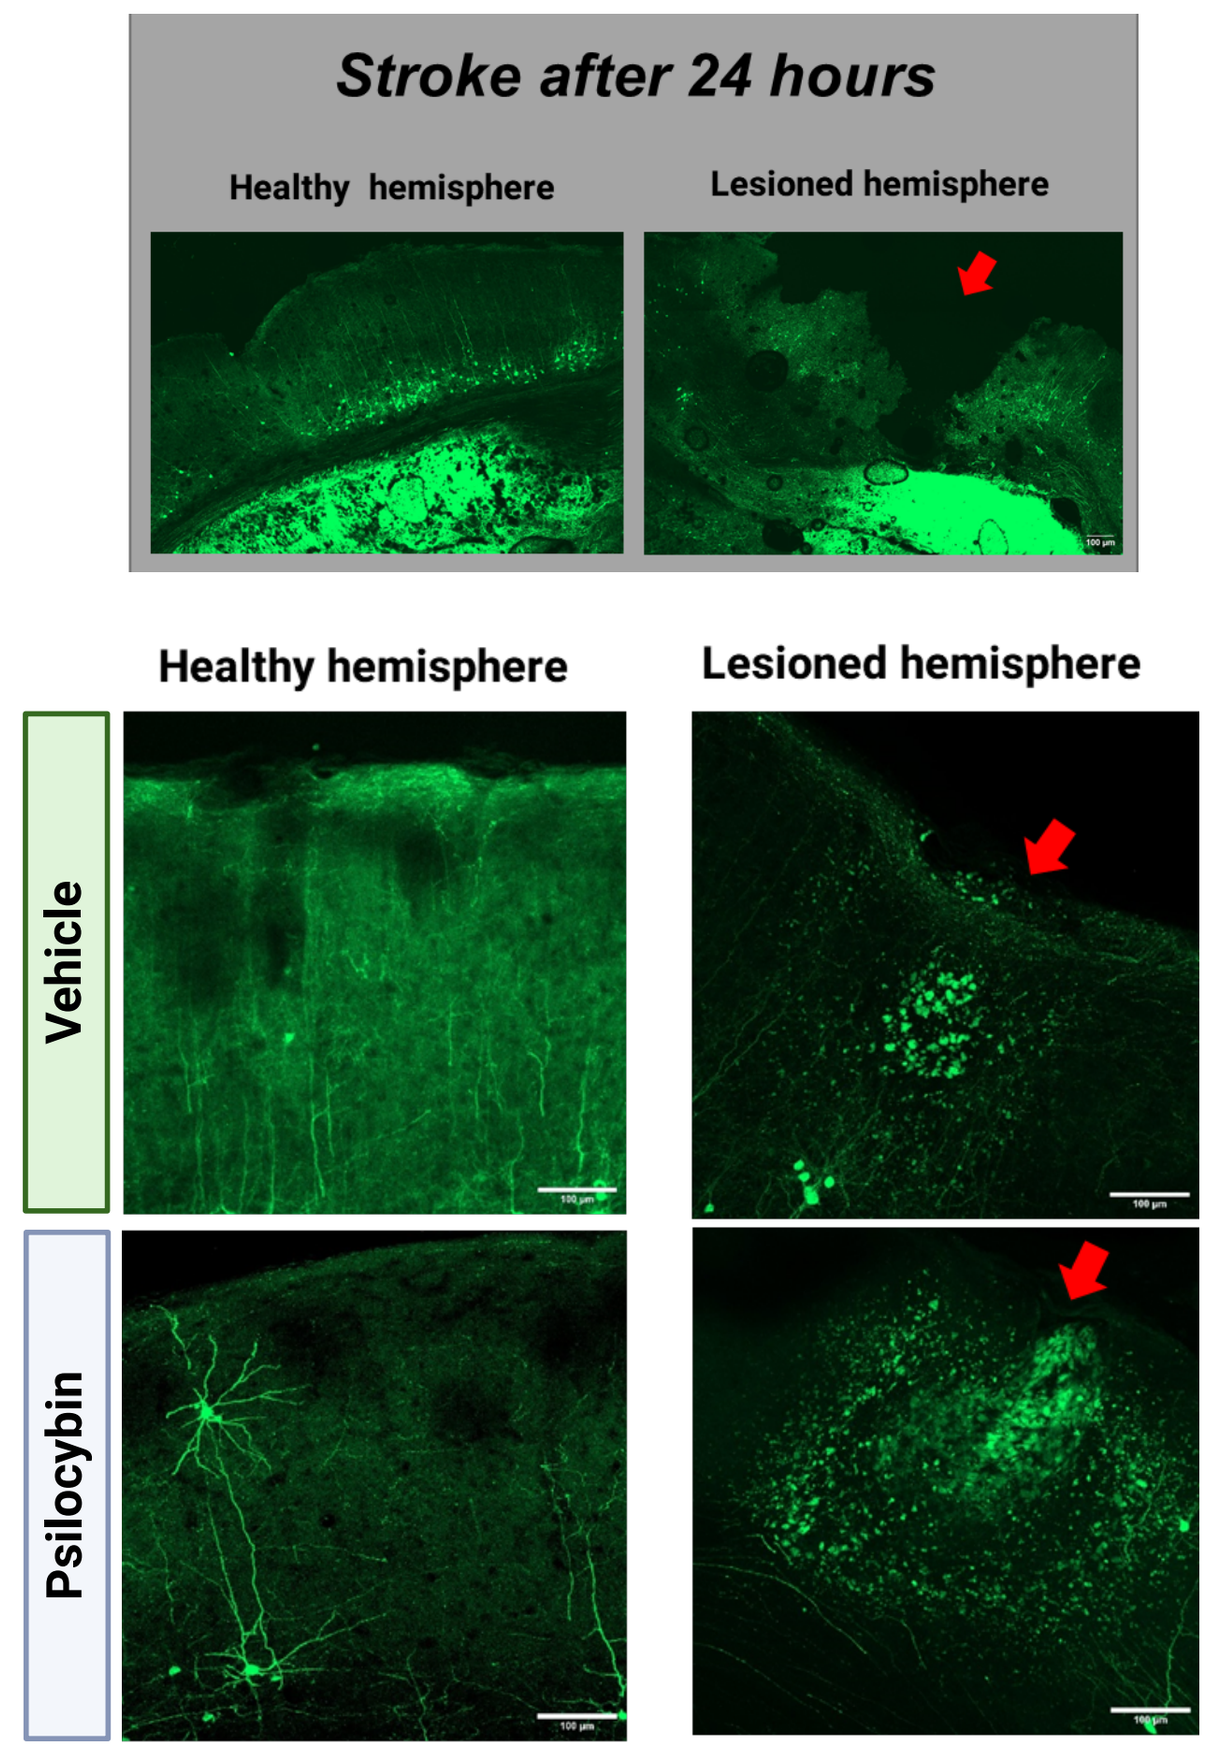
Stroke after 24 hours
Healthy hemisphere
Lesioned hemisphere
Vehicle
Psilocybin

Dendritic spines after 45 days of treatment

Psilocybin induces an increasing in spine density in the perilesion and distal area in both lesioned and healthy emispheres.

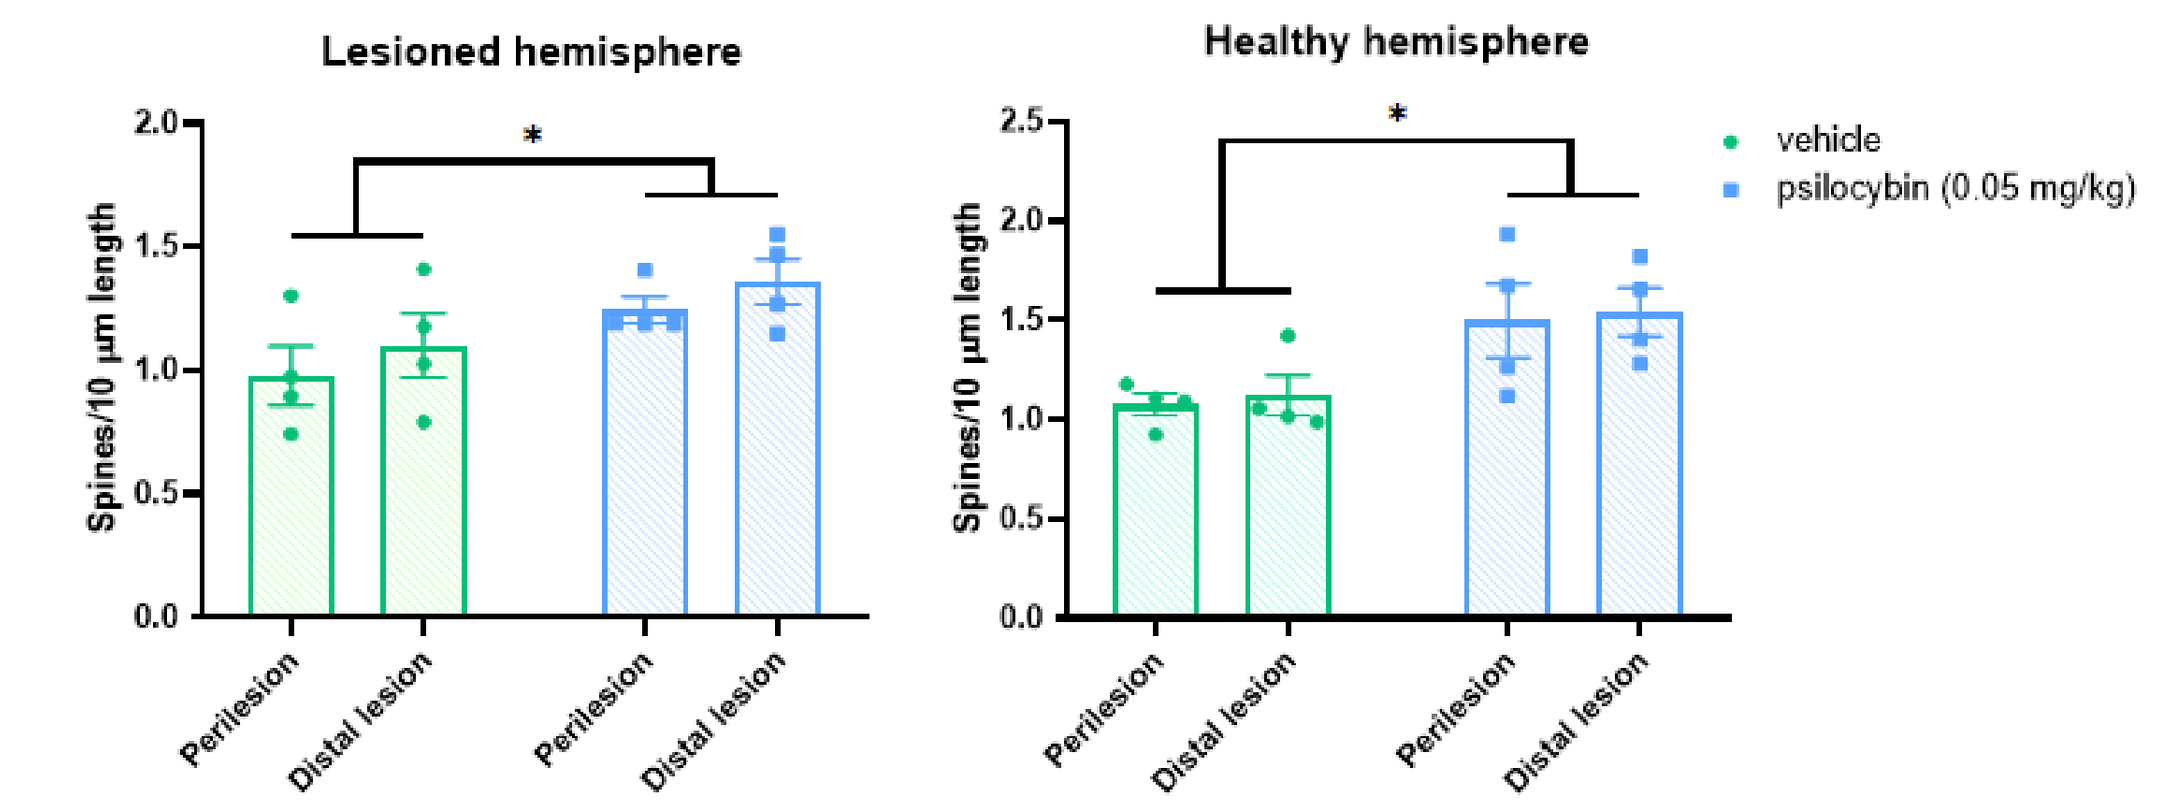
Lesioned hemisphere
Healthy hemisphere
Spines/10 µm length
Perilesion
Distal lesion
vehicle
psilocybin (0.05 mg/kg)
*

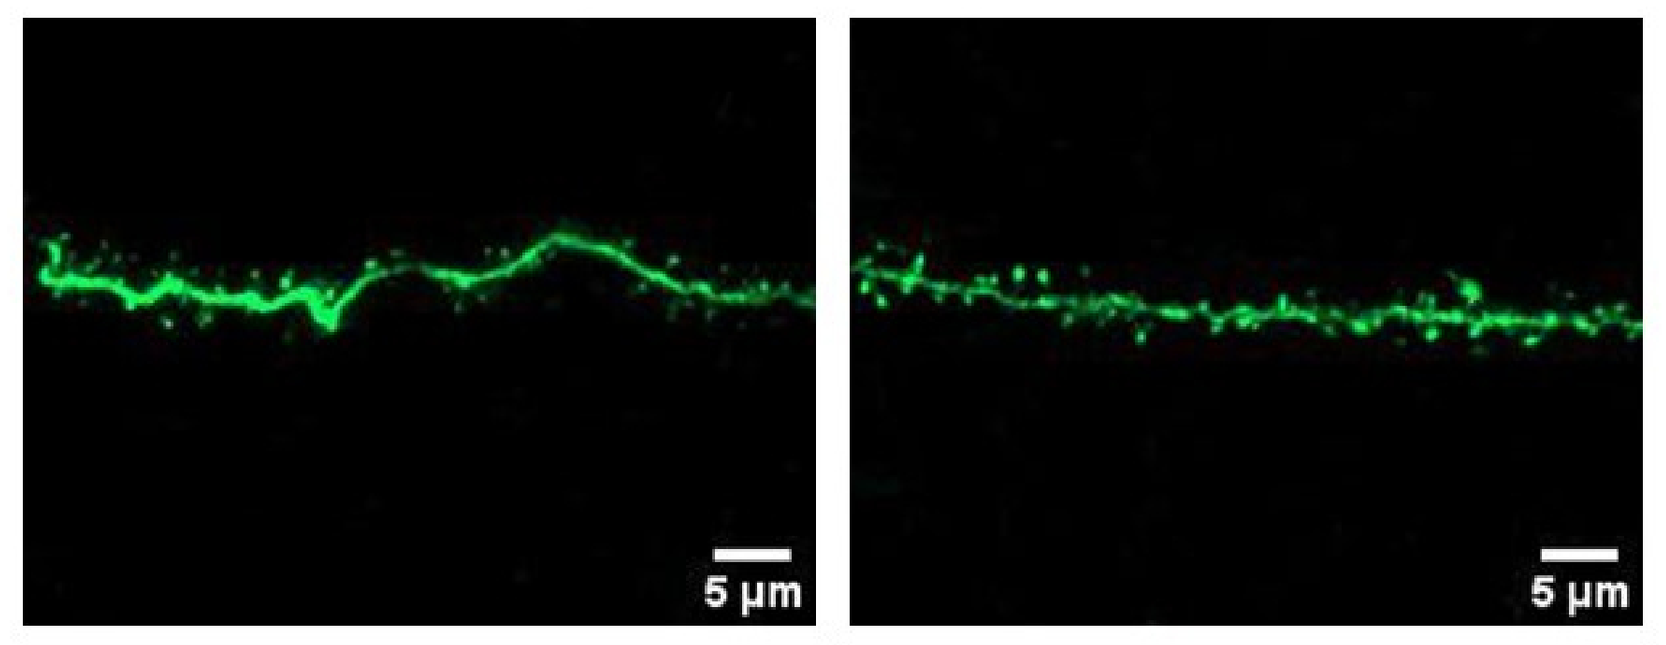
5 µm

Vehicle | Psilocybin

Images were acquired using a ZEISS LSM 800 Confocal Laser Scanning Microscope (Carl Zeiss, Jena, Germany) equipped with a Zeiss 63x/1.0– Plan-Apochromat oil objective using the ZEN Blue acquisition software.

CONCLUSION

Overall, these preliminary experiments suggest that a 45-day treatment with 0.05 mg/kg psilocybin, administered 24 hours after inducing a photothrombotic visual cortex stroke in mice, is well tolerated.
The treatment appears to improve tasks of visual performance to levels comparable to non-stroke mice and enhances dendritic spines in both the peri-lesional and distal regions of the injury.

DISCLOSURES: This work was funded by Relmada Therapeutics. Drs. Comai, Barzon, De Martin, Nasini and Mattarei, are employed by or have received compensation from companies or institutions that received funding from Relmada Therapeutics. Drs. Manfredi and Pappagallo received consultant fees from Relmada Therapeutics, Inc. Drs. Comai, DeMartin and Mattarei have received grant support from MGGM LLC and consultant fees from Neuroarbor LLC. Drs. Pappagallo and Manfredi are co-inventors of technology related to psilocybin. M. Cambiaghi: None.

REFERENCES

- Cambiaghi M, Cherchi L, Comai S. Photothrombotic Mouse Models for the Study of Melatonin as a Therapeutic Tool After Ischemic Stroke. *Methods Mol Biol.* 2022;2550:433-441. doi:10.1007/978-1-0716-2593-4_42
- Kerkhoff G. Neurovisual rehabilitation: recent developments and future directions. *J Neurol Neurosurg Psychiatry.* 2000;68(6):691-706. doi:10.1136/jnnp.68.6.691
- Ly C, Greb AC, Cameron LP, et al. Psychedelics Promote Structural and Functional Neural Plasticity. *Cell Rep.* 2018;23(11):3170-3182. doi:10.1016/j.celrep.2018.05.022

Created with BioRender Poster Builder