Effect of REL-1017 (esmethadone; d-methadone) on Cholesterol, Triglycerides, PCSK9, and hs-CRP in Patients with Major Depressive Disorder

Nicola Ferri [1];Marco Pappagallo [2,3]; Sheetal Patel [2]; Franco Folli [4]; Sara De Martin[5]; Maria Giovanna Lupo [5]; Sergio Traversa [2]; Paolo L. Manfredi [2]

[1] Department of Medicine, University of Padova, Padova, Italy; [2] Relmada Therapeutics, New York, NY, USA; [3] Department of Anesthesiology, Albert Einstein College of Medicine, Bronx, NY, USA; [4] University of Milano School of Medicine, Milano, Italy; [5] Department of Pharmaceutical and Pharmacological Sciences, University of Padova, Padova, Italy

INTRODUCTION

- Atherosclerotic cardiovascular disease (ASCVD) is the leading cause of morbidity and mortality in developed countries[1].
- The risk of clinical events will increase with cumulative exposure over the years of LDL cholesterol (LDL-C), underlining the importance of maintaining appropriate cholesterol levels overtime. The fundamental role of LDL-C in the pathogenesis of atherosclerosis, underscored by ample epidemiological and genetic evidence[1], suggests that determination of potential effects of drugs under clinical development on lipid metabolism may be prudent.
- Several classes of drugs have shown to alter lipid profile, including atypical antipsychotics in use for the treatment of major depressive disorder (MDD), such as olanzapine and risperidone[2]

OBJECTIVE

- We evaluated the effect of esmethadone, an N-methyl-D-aspartate receptor (NMDAR) antagonist under clinical development for the treatment of MDD on lipid metabolism and inflammatory markers.

METHODS

- Phase 2 study of esmethadone, 25 and 50 mg, in patients with Major Depressive Disorder (MDD).

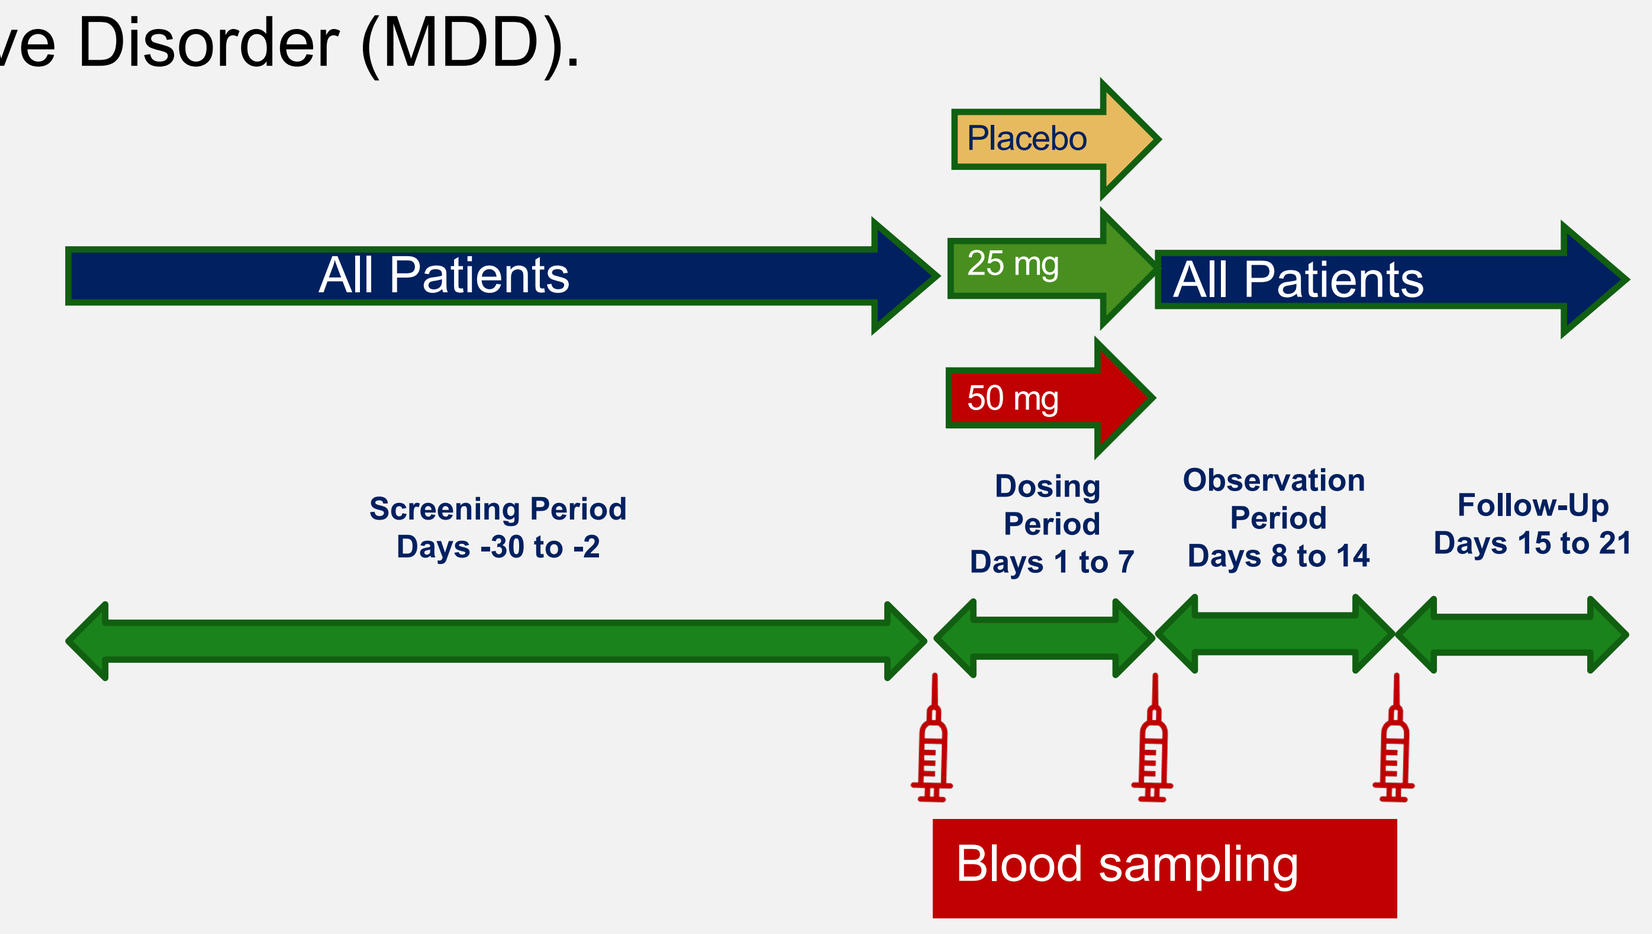
Placebo
All Patients
25 mg
All Patients
50 mg
Screening Period Days -30 to -2
Dosing Period Days 1 to 7
Observation Period Days 8 to 14
Follow-Up Days 15 to 21
Blood sampling

- **Blood sampling and ELISA**

Plasma samples collected on day 1 (baseline), day 7 (under treatment), and day 14 (1 week of wash-out) of patients with MDD treated with placebo (n=21), esmethadone 25 mg (n=16), and esmethadone 50 mg (n=19) were analyzed.

Plasma PCSK9 levels were measured in plasma aliquots using a commercial ELISA kit (R&D Systems, MN) able to identify both free and LDL-R-bound PCSK9. Samples were diluted 1:20 in accordance with the manufacturer's instructions, and incubated onto a microplate, which was pre-coated with a specific monoclonal antibody for the human PCSK9. A four-parameter logistic curve-fit was used to obtain sample concentrations (minimum detectable concentration: 0.219 ng/mL). The intra- and inter-assay coefficients of variability (CVs) were 5.4±1.2% and 4.8±1.0%, respectively.

High-sensitivity CRP (hs-CRP) levels were measured in plasma aliquots using a commercial ELISA kit (apDia bvba, Raadsherenstraat 3, 2300 Turnhout, Belgium). The intra- and inter-assay coefficients of variability (CVs) were 5.1±1.6% and 6.1±0.3%, respectively.

- **Quantifications of total cholesterol and triglycerides**

For total cholesterol (Cholesterol CP, ABX Pentra, cod. A11A01634) and triglycerides (Triglycerides CP, ABX Pentra, cod. A11A01640) evaluation 10 μL of plasma samples were used according to manufacturer's instruction. The colorimetric products by enzymatic reactions were recorded at 570 nm with Victor Nivo spectrophotometer (PerkinElmer, Waltham, Massachusetts, USA). Limit of detection for total cholesterol assay was 0.09 mmol/L (3.48 mg/mL), whilst intra- and inter-assay accuracies were 0.65% and 2.7%, respectively. Limit of detection for triglyceride assay was 0.08 mmol/L (7 mg/mL), while intra- and inter-assay accuracies were 1.5% and 1.63%, respectively.

RESULTS

Figure 1. Total cholesterol and triglyceride levels of placebo, esmethadone 25mg, and 50mg treated patients. A) all study group; B) patients not treated with statins

No significant changes on total cholesterol and triglycerides levels were observed in response to esmethadone treatment at both 25 mg and 50 mg when compared to placebo and between different time points **(Table 1 and Figure 1).**

Proprotein convertase subtilisin/kexin type 9 (PCSK9) represents the most important pharmacological target for reducing the LDL-C levels and our research group has recently discovered its possible involvement in the link between depression, insulin resistance, and cardiovascular risk factors [3]

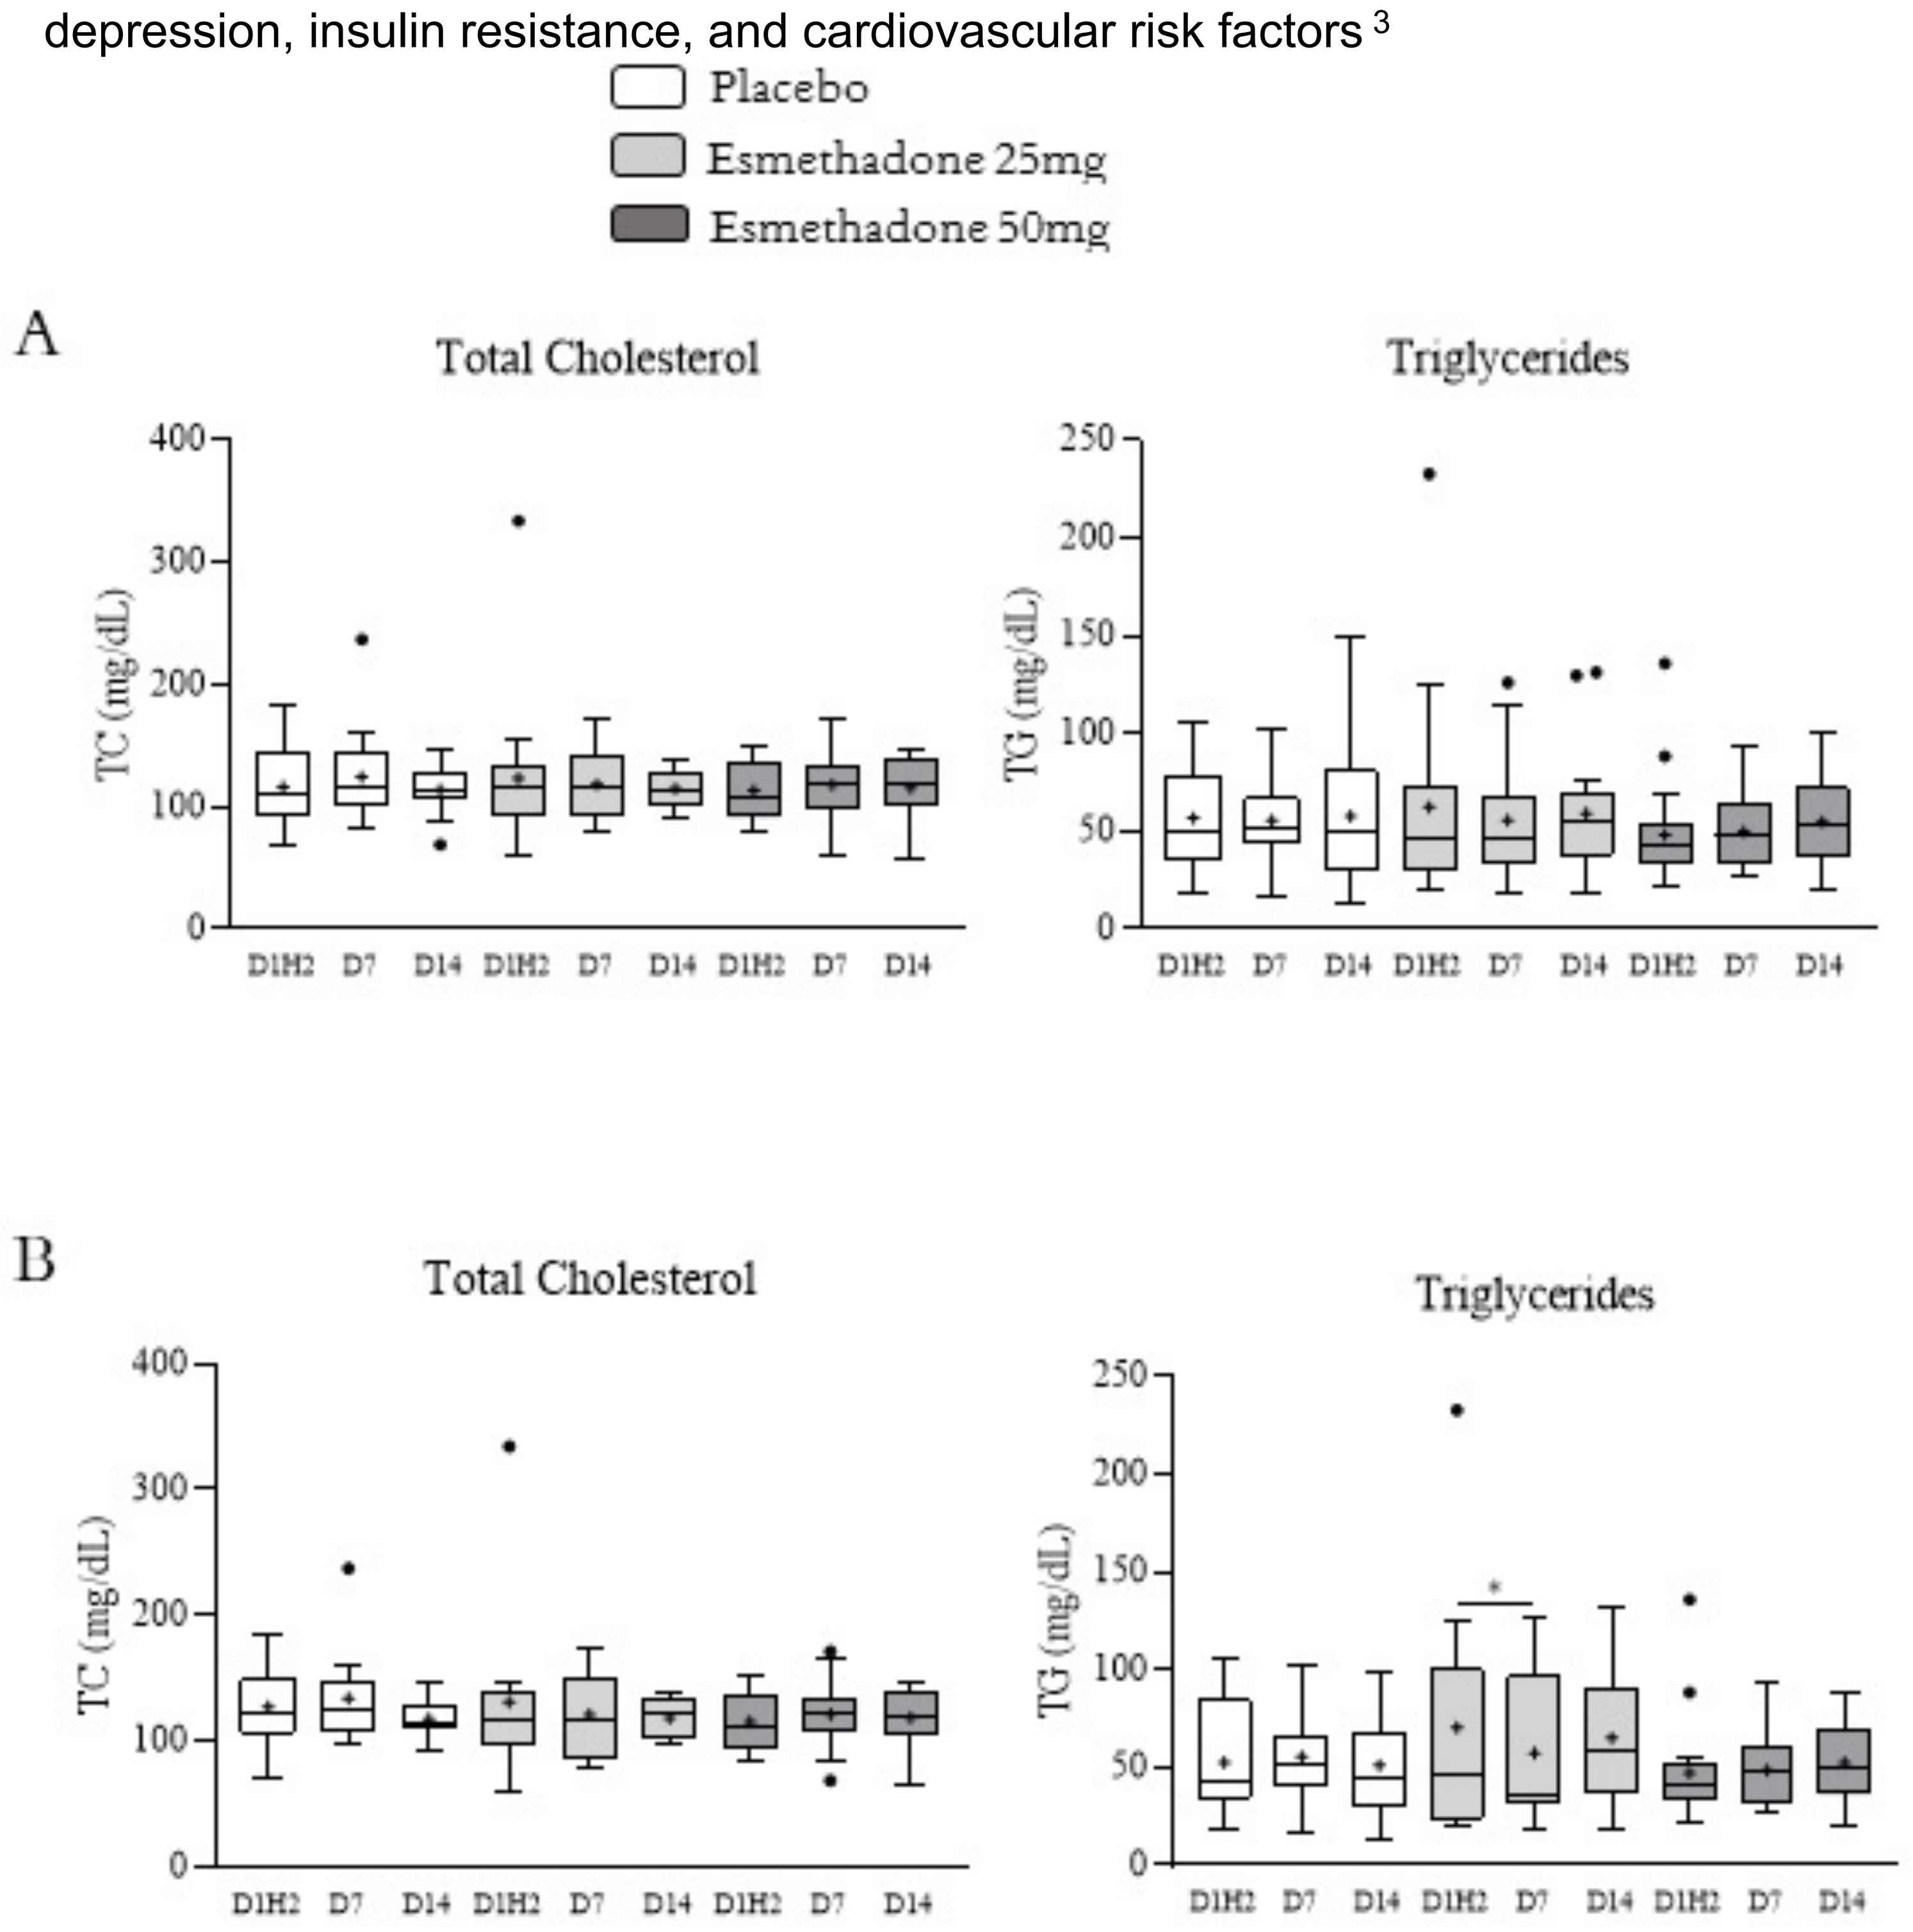
Placebo
Esmethadone 25mg
Esmethadone 50mg
A
Total Cholesterol
Triglycerides
TC (mg/dL)
TG (mg/dL)
B
Total Cholesterol
Triglycerides

Figure 2. PCSK9 levels of placebo, esmethadone 25mg, and 50mg treated patients. A) all study group; B) patients not treated with statins.

*p<0.05; **p<0.01; ***p<0.001 Student's t-tests.

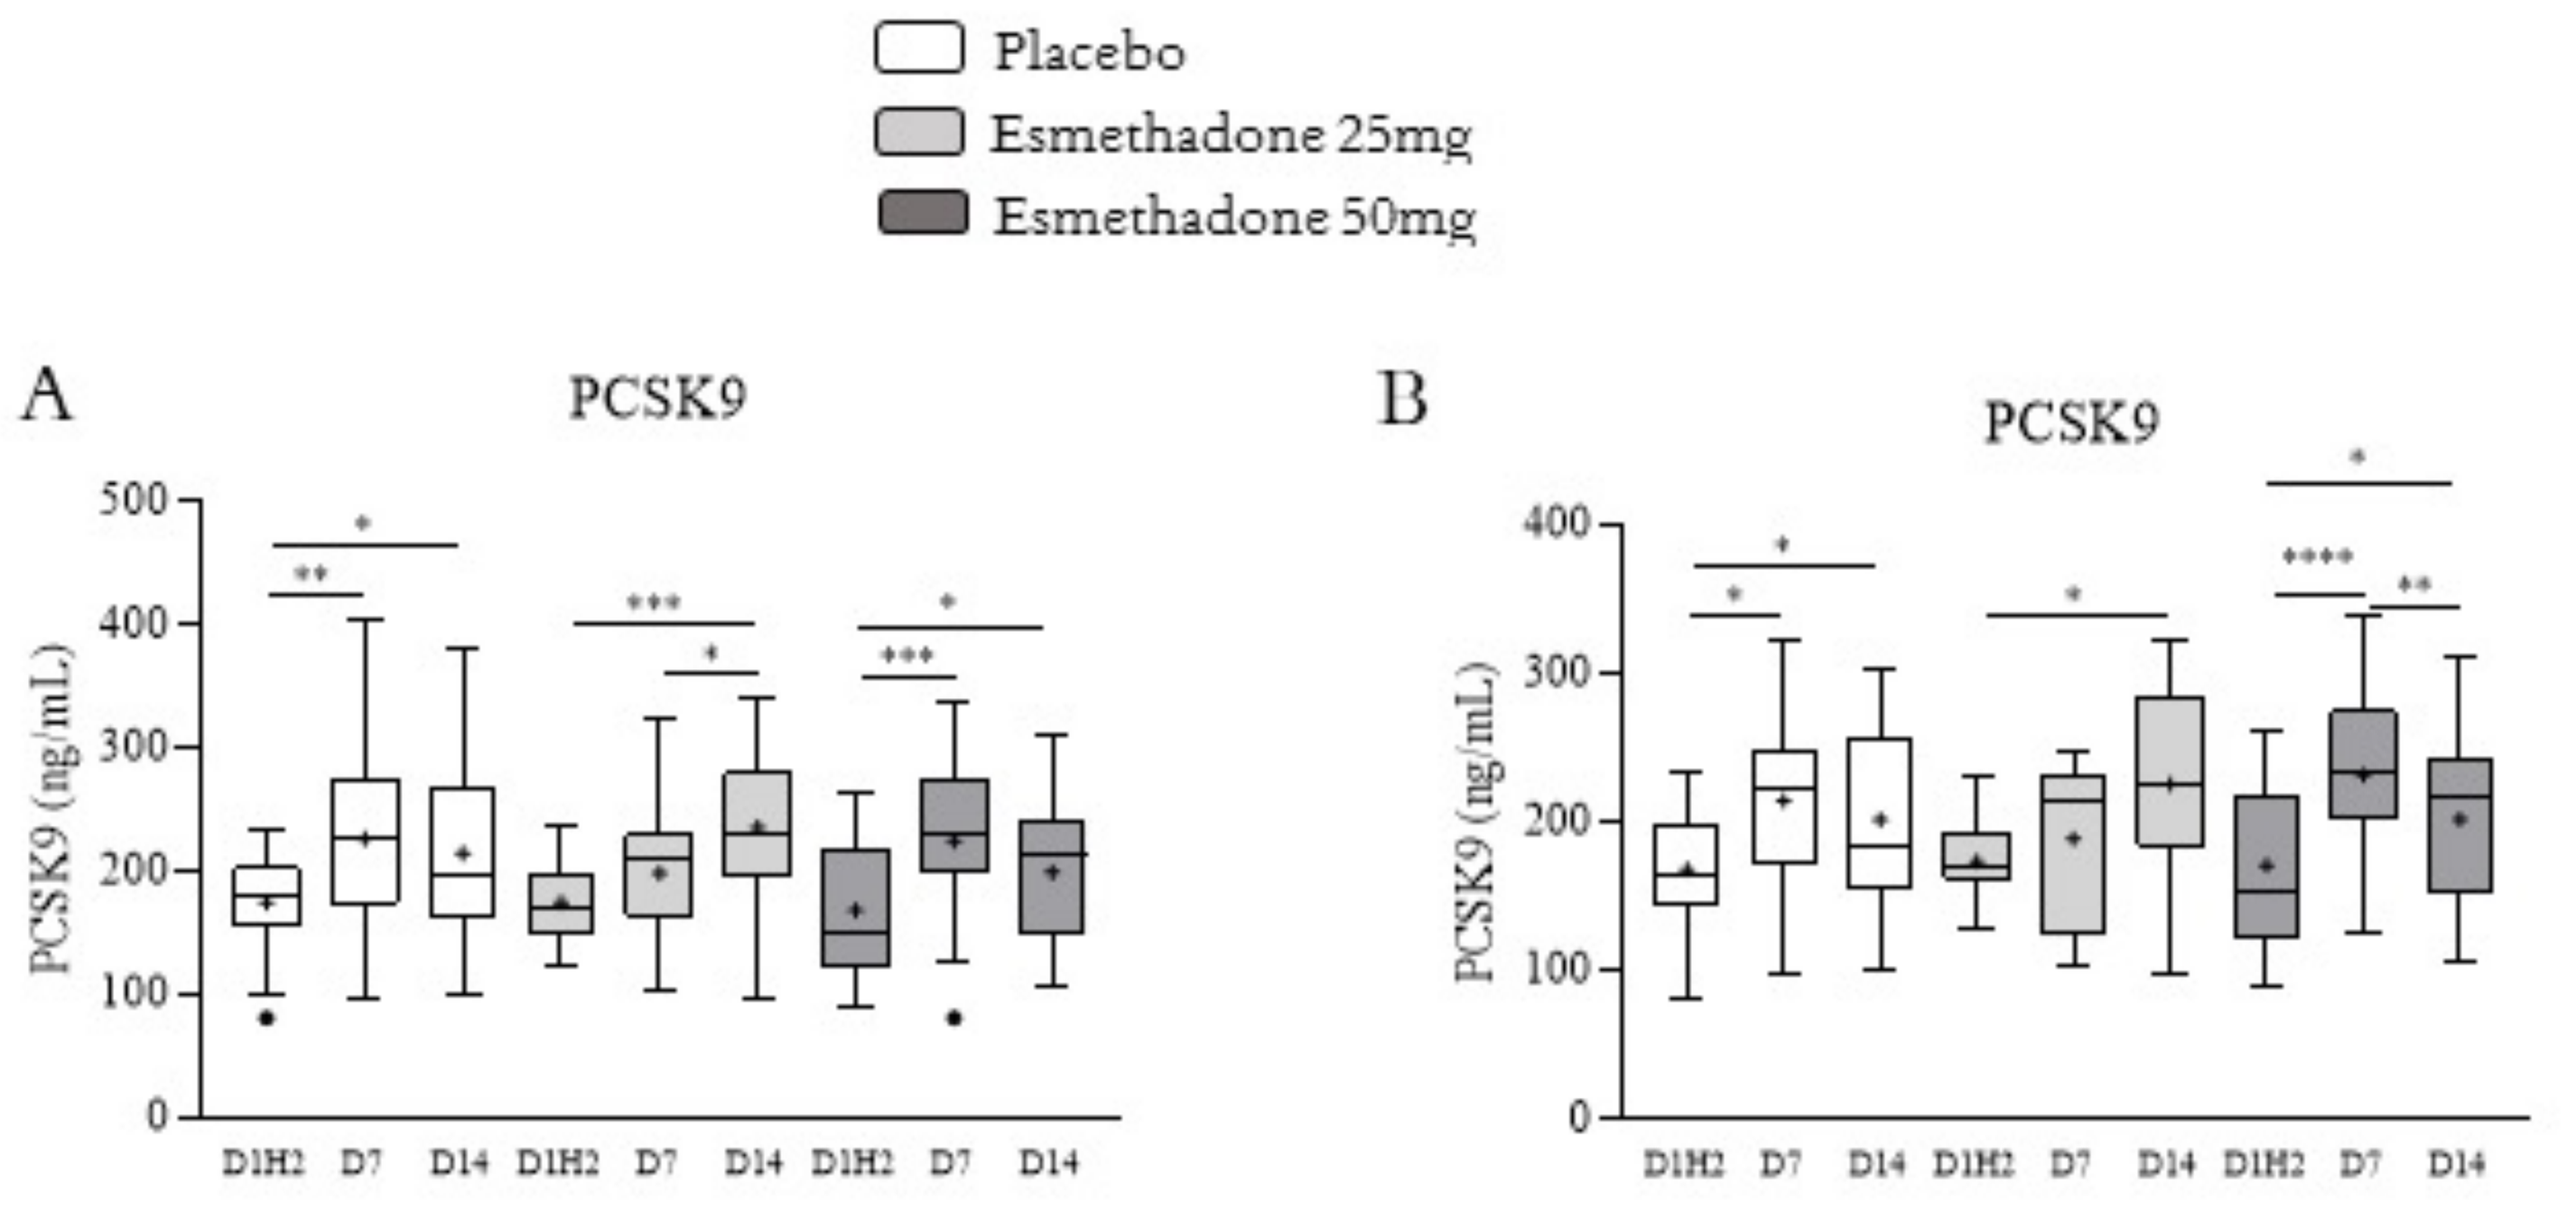
Placebo
Esmethadone 25mg
Esmethadone 50mg
A
PCSK9
B
PCSK9
PCSK9 (ng/mL)

Table 1. Baseline total cholesterol and triglycerides levels of patients treated with placebo, esmethadone 25 mg, and 50 mg.

	Placebo (n=21)	Esmethadone 25 mg (n=16)	Esmethadone 50 mg (n=19)
Cholesterol (mg/dL)	117.8±30.8	123.7±59.3	113.9±22.9
Triglycerides (mg/dL)	57.0±25.6	62.7±52.1	48.5±25.3
	Placebo (no statins; n=15)	**Esmethadone 25 mg (no statins; n=10)**	**Esmethadone 50 mg (no statins; n=18)**
Cholesterol (mg/dL)	127.5±28.6	131.0±71.7	115.5±22.1
Triglycerides (mg/dL)	52.9±27.6	70.6±63.3	47.4±25.4

The total cholesterol levels at baseline did not differ between the three different groups and the mean of all patients recruited in the study was 117.7±38.6 mg/dL. According to the current guidelines for the management of dyslipidemias, some of the patients were under statin treatment. Indeed, 6 out of 21, 6 out of 16, and 1 out of 19, were under statin therapy in the placebo, esmethadone 25mg, and 50mg group, respectively.

Figure 3. hs-CRP levels of placebo, esmethadone 25mg, and 50mg treated patients. A) all study group; B) patients not treated with statins.

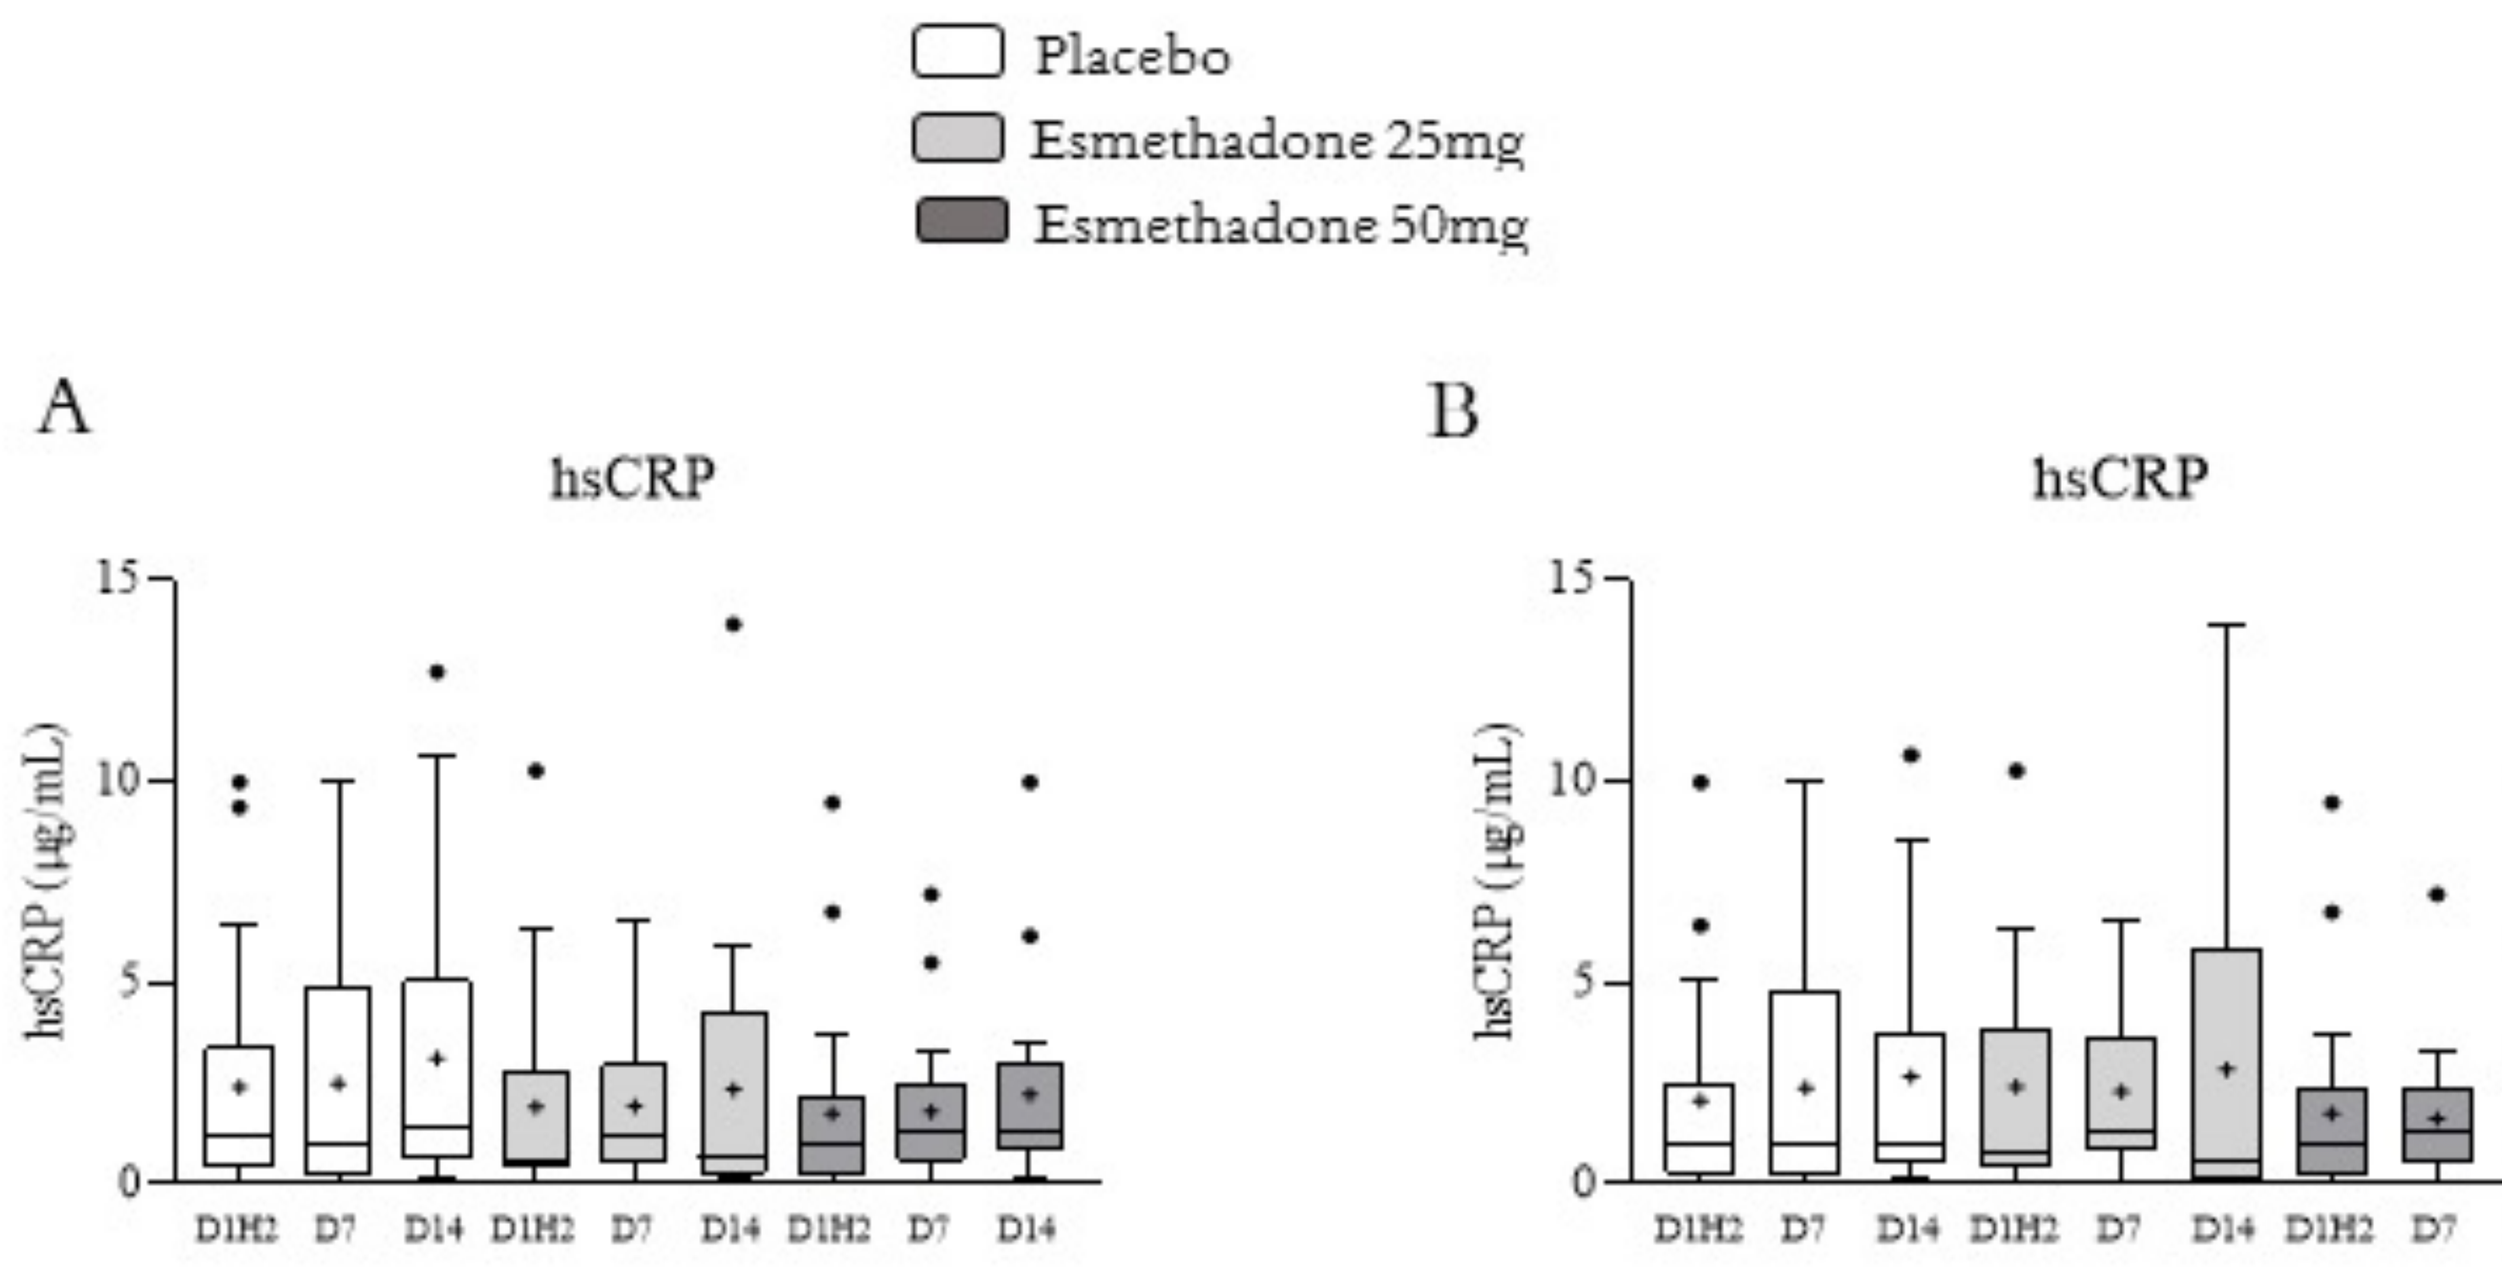
Placebo
Esmethadone 25mg
Esmethadone 50mg
A
hsCRP
B
hsCRP
hsCRP (μg/mL)

C reactive protein (CRP) is a liver-derived acute phase protein associated with inflammation. High sensitivity (hs)-CRP>2 mg/L associates with major CV risk markers, such as elevated LDL-C, as well as with the progression of CV lesions.

30% of the patients had hs-CRP plasma levels higher than 2 mg/L, thus potentially associated to higher incidence of CV events. Seven days of oral daily treatment with 25 mg or 50 mg esmethadone did not alter hs-CRP plasma levels.

CONCLUSIONS

- Considering the relationship between CVD and depression, it is prudent to study the effect of antidepressant drugs on major risk factors for cardiovascular disease, such as cholesterol, triglycerides, PCSK9, and certain inflammatory markers (hs-CRP).
- We measured these parameters at baseline, after 7 days of treatment (placebo, 25mg, and 50mg of esmethadone), and after an additional 7 days of wash out from esmethadone treatment.
- This set of patients were under treatment with additional antidepressant drugs and some patients were receiving hypocholesterolemic drugs (statins).
- Our analyses showed that esmethadone did not significantly alter lipid parameters, PCSK9, and hs-CRP, thus suggesting the absence of any detrimental effect on ASCVD risk.

REFERENCES

1. Ference BA, *et al*. Impact of Lipids on Cardiovascular Health: JACC Health Promotion Series. J Am Coll Cardiol 2018;72(10):1141-1156.
2. Perez-Iglesias R, *et al*. A 12-week randomized clinical trial to evaluate metabolic changes in drug-naive, first-episode psychosis patients treated with haloperidol, olanzapine, or risperidone.
3. J Clin Psychiatry 2007;68(11):1733-40.Macchi C, Favero C, Ceresa A, Vigna L, Conti DM, Pesatori AC, et al. Depression and cardiovascular risk - association among Beck depression inventory, PCSK9 levels and insulin resistance. Cardiovascular Diabetology 2020.

DISCLOSURES

This research was sponsored by Relmada Therapeutics, Inc. Drs. Folli, Pappagallo, and Manfredi are paid consultants of Relmada Therapeutics. Drs. Patel and Traversa are employees of Relmada Therapeutics. Drs. Ferri, De Martin, and Lupo are employed or have received fees from companies or Universities that have received payments or grants from Relmada. Dr. Manfredi is an inventor on esmethadone patents and other patents and patent applications.

NEI Annual Congress 2021

Corresponding Author: Paolo L. Manfredi, M.D. Email: pmanfredi@relmada.com