1067-P, American Diabetes Association, 13-17th June 2014, San Francisco, USA

Imeglimin Decreases Hepatic Glucose Production through a Unique Mitochondrial Mechanism of Action

Guillaume Vial[1,2] PhD; Frédéric Lamarche[3,4] PhD; Anne-Laure Borel[5] PhD ; Dominique Detaille[6] PhD; Cécile Cottet-Rousselle[3,4]; Sophie Hallakou-Bozec[7] PhD; Sebastien Bolze[7] PhD; Éric Fontaine[3,4,5] PU-PH

[1]INSERM U1060, Laboratoire CarMeN, Faculté de médecine Lyon-Sud, France; [2]European Center For Nutrition and Health, Centre Hospitalier Lyon Sud, France; [3]INSERM U1055, Laboratoire de Bioénergétique Fondamentale et Appliquée (LBFA) et SFR Biologie Environnementale et Systémique (BEeSy), Grenoble, France; [4]Université de Grenoble F-38041 Grenoble cedex 9; [5]University Hospital, Grenoble, France; [6]INSERM U1045 – Cardio-Thoracic Research Centre – and Rhythmology and Heart Modeling Institute (LIRYC), Bordeaux University, Bordeaux, France; [7]Poxel SA, Lyon, France

Abstract

Imeglimin (IME) is the first in a new class (Glimins) of oral glucose-lowering agents currently in phase IIb development. IME has been previously shown to decrease glucose production in hepatocytes. The aim of this study was to elucidate the underlying mechanism of action. As gluconeogenesis (GNG) is an energy consuming pathway controlled by ATP/ADP, we investigated this ratio as well as the upstream mitochondrial events (redox potential, membrane potential and respiration) in primary rat hepatocytes after an overnight incubation with IME or metformin (MET). IME and MET dose-dependently decreased glucose production (-56% vs. -90% at 1mM, $p<0.05$) with a concomitant marked decrease in ATP/ADP (-61% vs -78% at 1mM; $p<0.05$). IME-induced glucose production inhibition was not associated with an effect on mitochondrial respiration by contrast to MET which decreased it by 48% at 1mM due to the inhibition of mitochondrial complex1 (C1). Here we demonstrated that IME triggered a competitive C1 inhibition (same Vmax but Km increased 3-fold) whereas MET induced an uncompetitive C1 inhibition (Vmax decreased 1.4-fold and Km increased 4-fold). To overpass such a kinetic constraint, the redox potential (NADH/NAD, assessed by confocal microscopy) is increased and the mitochondrial membrane potential (assessed by confocal microscopy) is decreased. IME decreased ATP/ADP ratio without any decrease in inorganic phosphate levels leading to a reduction in the energy available per molecule of ATP (ΔG). As a consequence, this dramatically affects the activity of ATP consuming pathways such as GNG. We conclude that IME strongly decreased GNG and phosphate potential without inhibiting mitochondrial respiration. Glimins do not induce the mitochondrial respiration impairments observed with biguanides, and therefore have the advantage of avoiding lactic acidosis risk. This unique mechanism of action is different from the one described for MET and explains the excellent safety profile of IME.

Background

- Imeglimin is the first in a new tetrahydrotriazine-containing class of oral anti-diabetic agents, the Glimins.
- The efficacy of Imeglimin has been demonstrated in T2DM patients, both as mono and add-on (metformin and sitagliptin) therapies with no safety signals to date (600 subjects)[1–3].
- Imeglimin is currently in phase IIb clinical development (US/EU).
- Imeglimin regulates the mitochondrial bioenergetics and targets the 3 key organs (liver, muscle, pancreas) implicated in T2DM[4].
- Imeglimin has also been shown to increase insulin secretion in response to glucose (Imeglimin Increases Glucose-dependent Insulin Secretion and Improves Beta-Cell Function in Patients with Type 2 Diabetes - Pr M.Roden, ADA 2014, 120-OR).
- Imeglimin improves liver and muscle insulin sensitivity (Imeglimin Normalizes Glucose Tolerance and Insulin Sensitivity in Improving Mitochondrial Function in a High-Fat High-Sucrose Diet Mice Model – Dr S. Hallakou-Bozec, ADA 2014, 119-OR).

Objectives

To elucidate the underlying mechanisms by which Imeglimin decreases gluconeogenesis in primary rat hepatocytes in comparison to metformin.

Research Design and Methods

After an overnight exposure to the drugs (1mM), we compared the effects of Imeglimin and metformin in primary rat hepatocytes on several parameters; glucose production, ATP/ADP.Pi ratio, oxygen consumption rate, mitochondrial redox potential (NADH), and membrane potential (TMRM).

Results

Imeglimin and metformin decreased glucose production and ATP/ADP ratio

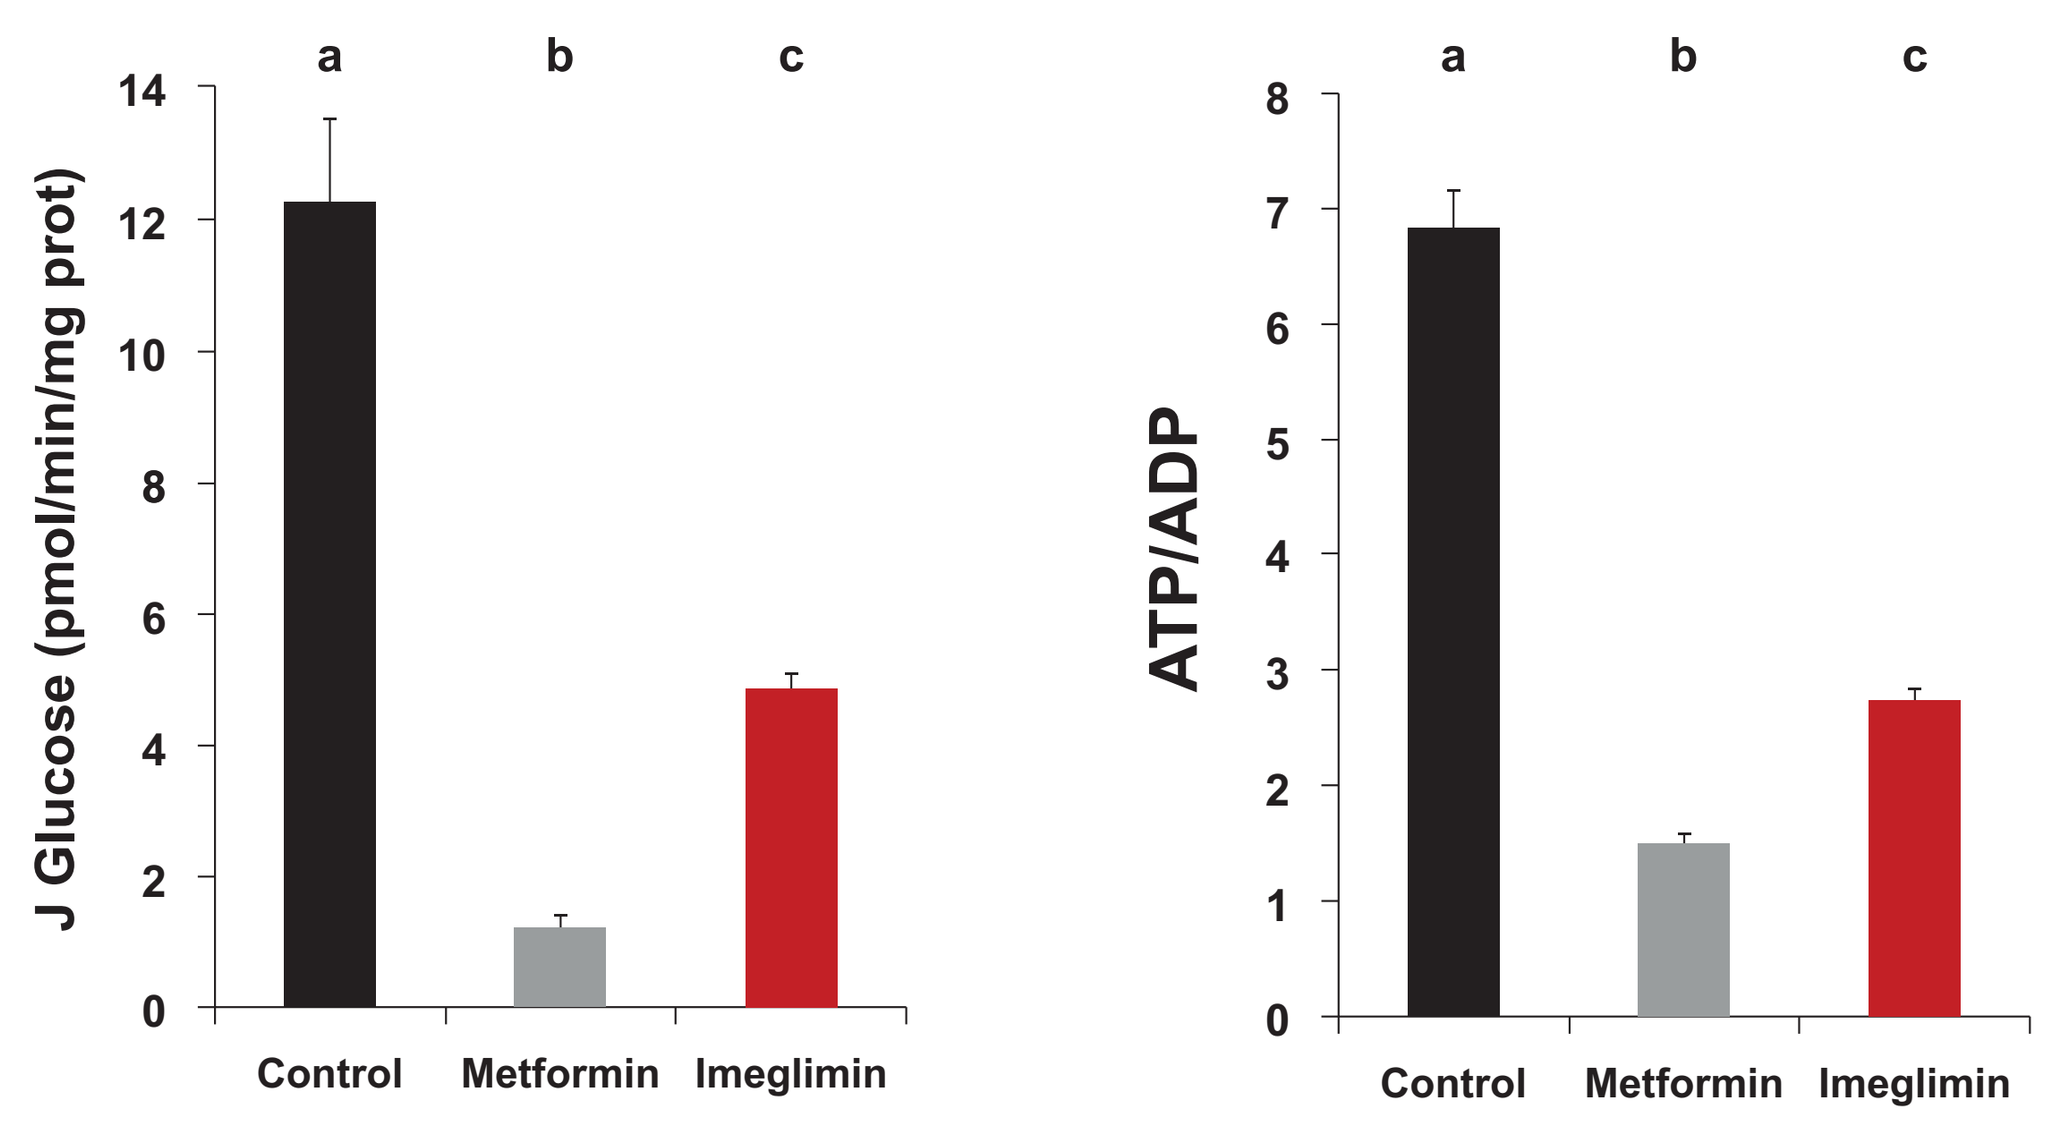
J Glucose (pmol/min/mg prot)
ATP/ADP
a
b
c
Control
Metformin
Imeglimin

a, b, c values not connected by the same letter are significantly different

► What are the mitochondrial events linked to these effects?

Contrary to metformin, Imeglimin did not affect mitochondrial respiration rate

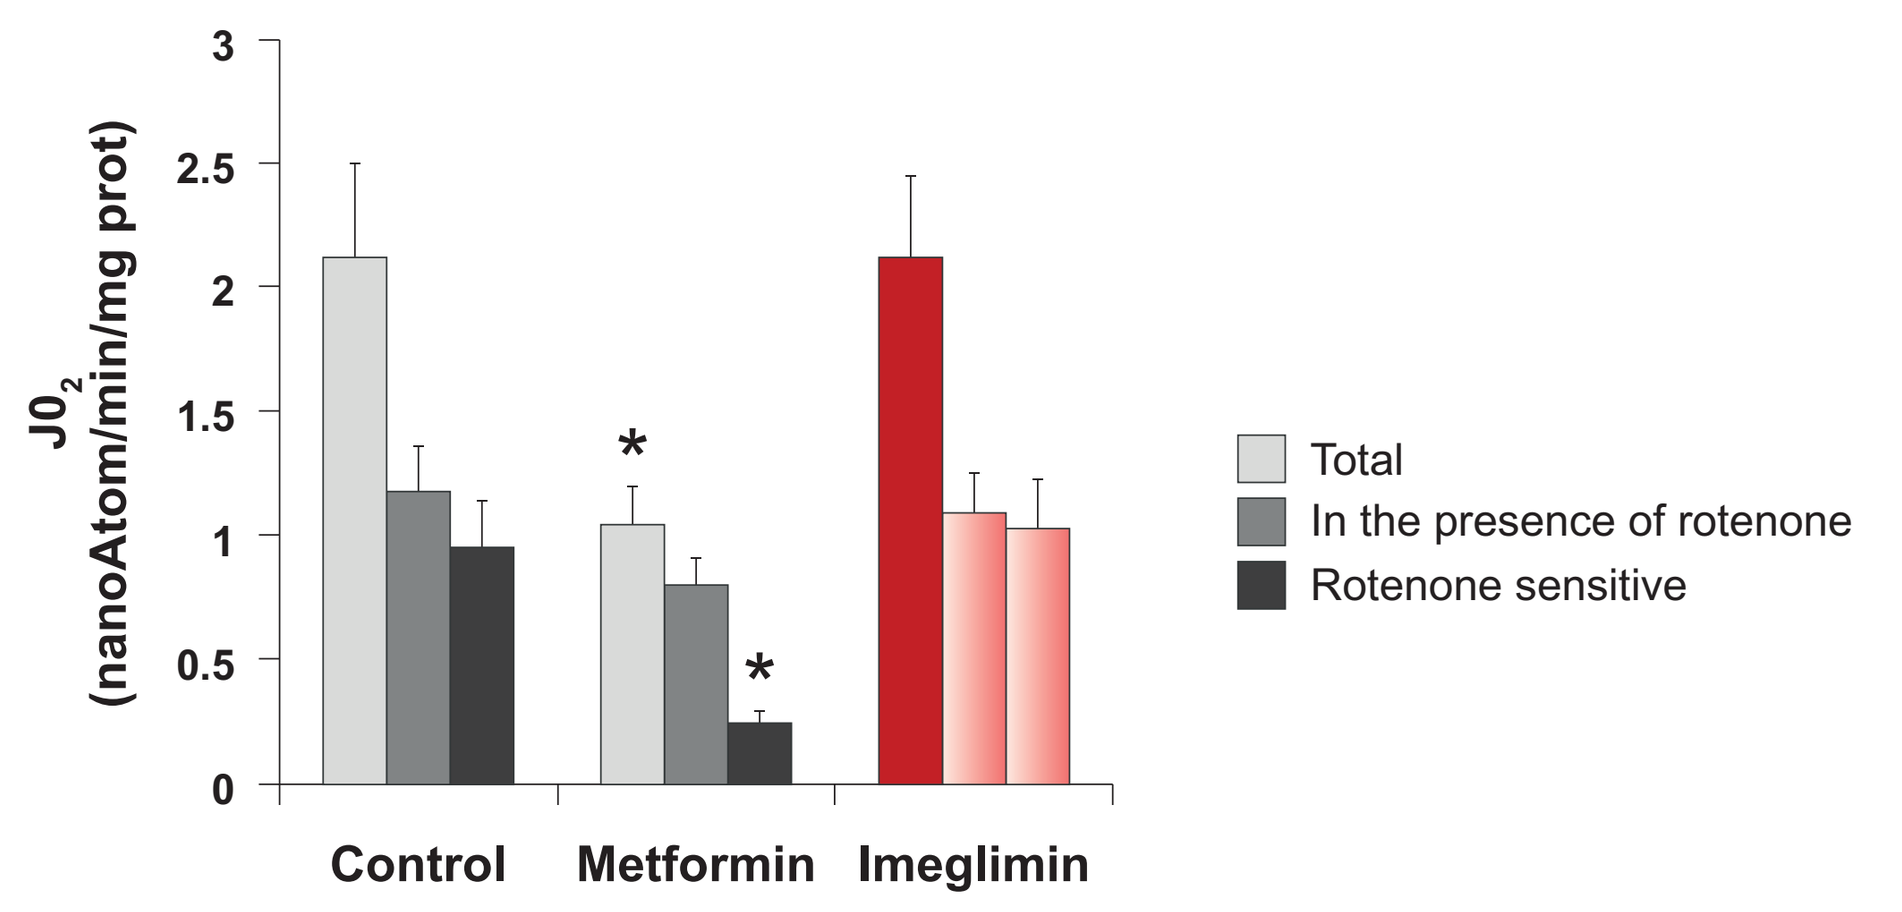
JO_2 (nanoAtom/min/mg prot)
Total
In the presence of rotenone
Rotenone sensitive
Control
Metformin
Imeglimin

*; p<0.05 vs respective control

- Metformin decreases rotenone sensitive respiration which is consistent with its well-known inhibitory effect on complex I.
- Imeglimin exhibits no effect on mitochondrial respiration.

Mitochondrial respiration is linked to the turnover of ATP (ATP flux).

► How can we explain Imeglimin decrease in ATP/ADP ratio in the presence of a normal oxygen consumption?

Three forces drive mitochondrial respiration and phosphorylation rate: redox potential; proton-motive force; and phosphate potential.

► What's happening on these parameters?

Imeglimin has an innovative effect on the respiratory chain

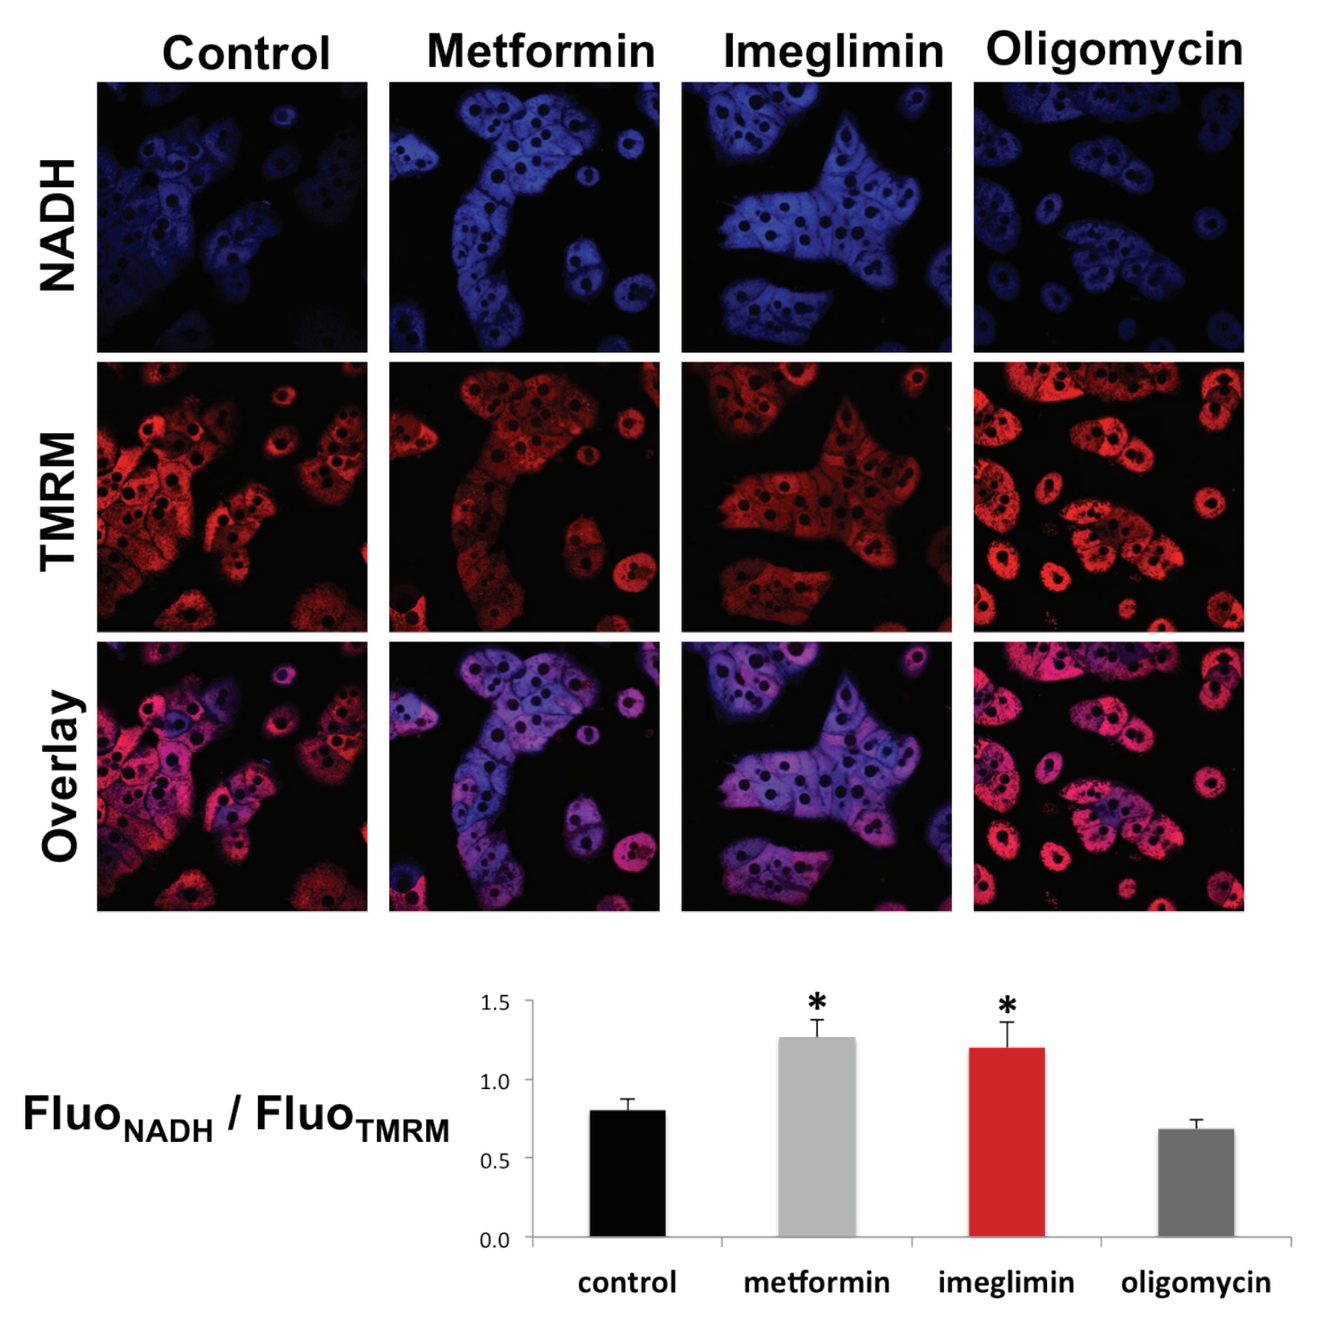
Control
Metformin
Imeglimin
Oligomycin
NADH
TMRM
Overlay
$Fluo_{NADH}$ / $Fluo_{TMRM}$
control
metformin
imeglimin
oligomycin

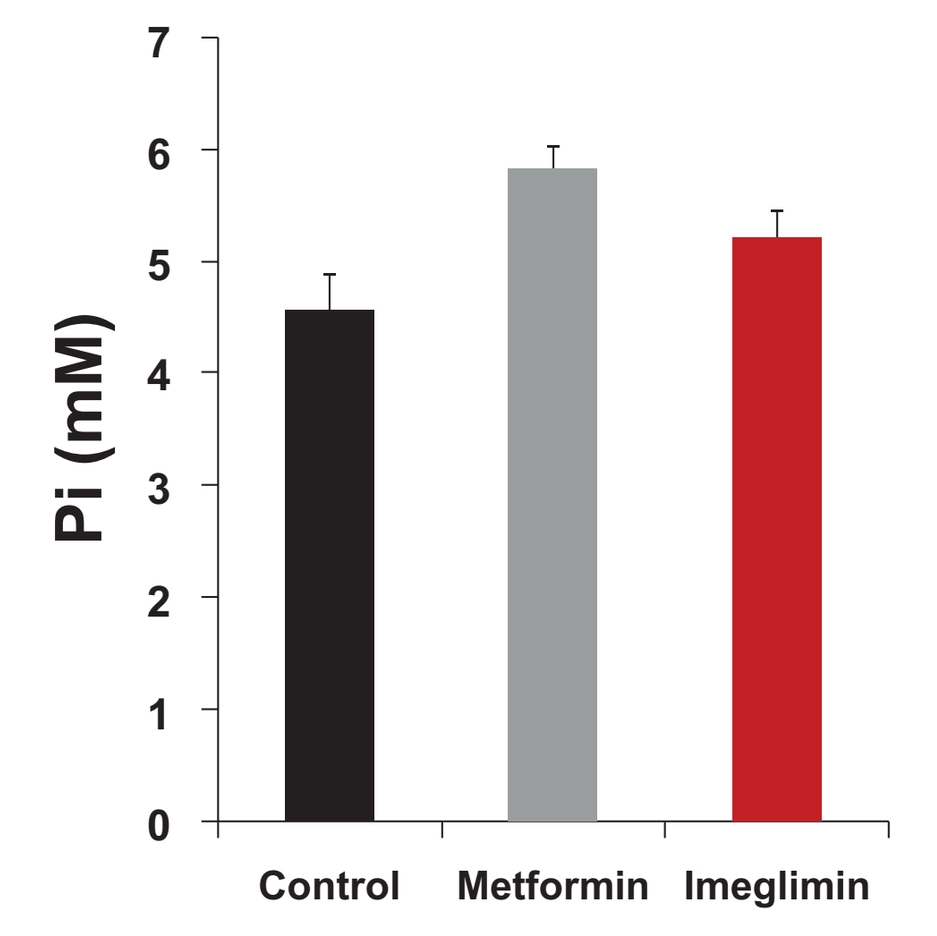
Pi (mM)
Control
Metformin
Imeglimin

ΔG ATP kJ/mol
Control: -49.50 kJ/mol
Metformin: -44.93 kJ/mol
Imeglimin: -46.72 kJ/mol

- Imeglimin and metformin increase redox potential (NADH) and decrease membrane potential (TMRM), as well as phosphate potential (estimated by ATP/ADP.Pi).
- Imeglimin innovatively affects the respiratory chain by inducing an increase in redox potential and a decrease in membrane potential without modifying respiration.

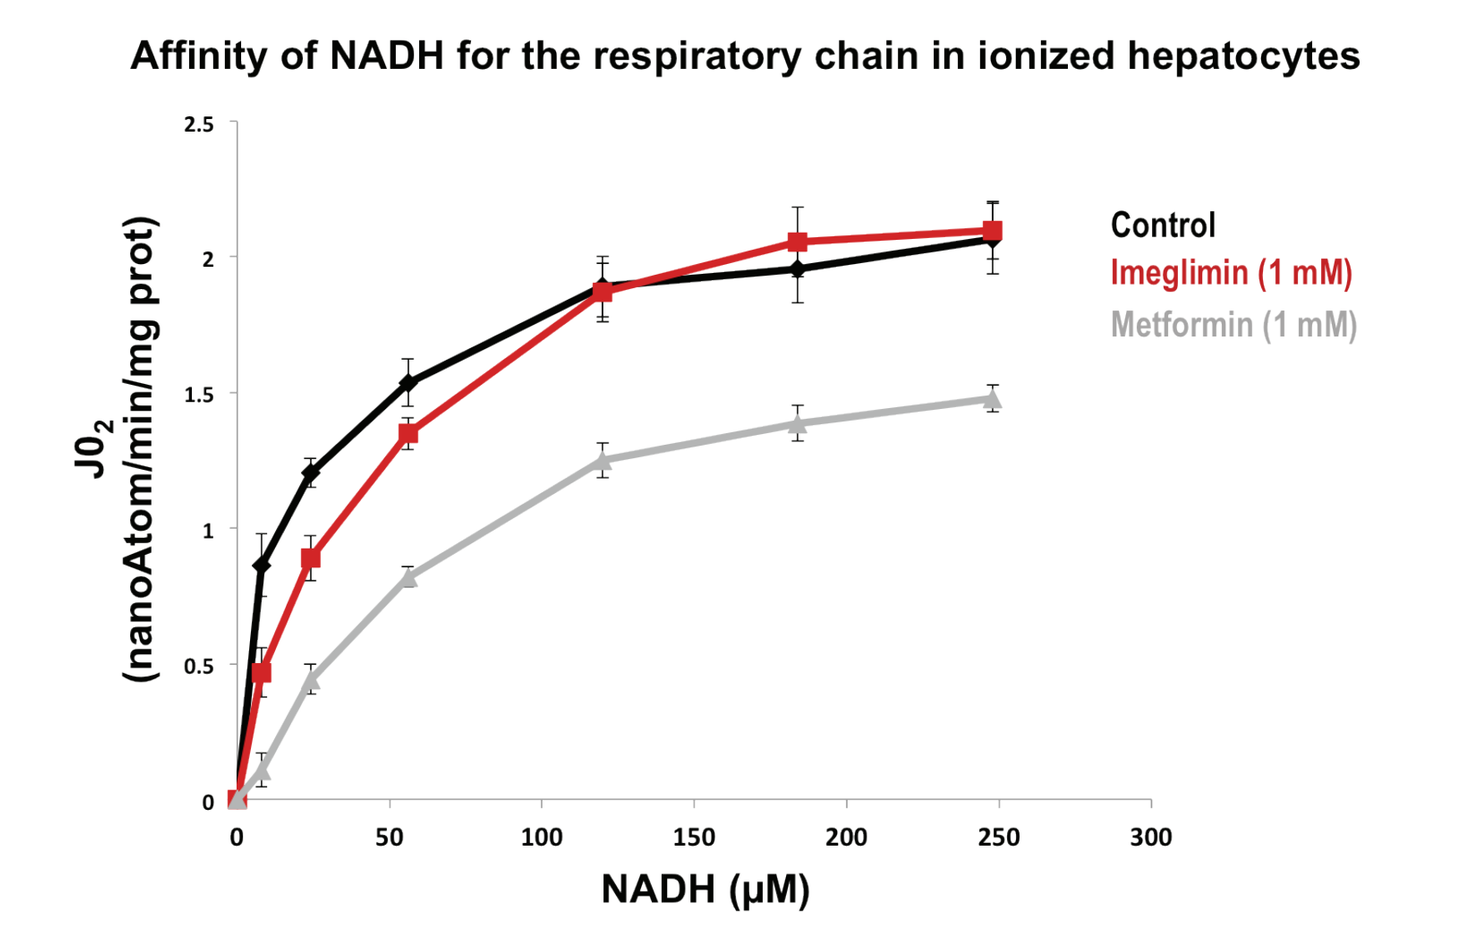
Affinity of NADH for the respiratory chain in ionized hepatocytes
JO_2 (nanoAtom/min/mg prot)
NADH (µM)
Control
Imeglimin (1 mM)
Metformin (1 mM)

Hepatocytes were exposed to an osmotic shock and then incubated in the presence of the indicated concentration of NADH. Rotenone-sensitive oxygen consumption rate was measured (JO_2) before and after the addition of rotenone.

- Imeglimin acts as a competitive inhibitor of the respiratory chain.
 - o The competitive inhibition on the respiratory chain can be overcome by both an increase in redox potential (NADH) and a decrease in the proton-motive force, the latter being responsible for a thermodynamic constraint on the ATP synthesis (ΔG ATP).
- Metformin acts as an uncompetitive inhibitor of the respiratory chain.
 - o The uncompetitive inhibition on the respiratory chain leads to a decrease in respiratory flux, membrane potential, phosphate potential and thereby a reduction in ATP synthesis.

Conclusion

Imeglimin inhibits gluconeogenesis by inducing a thermodynamic constraint on ATP synthesis but, contrary to metformin, without affecting respiratory flux thereby preventing Imeglimin from inducing lactic acidosis.

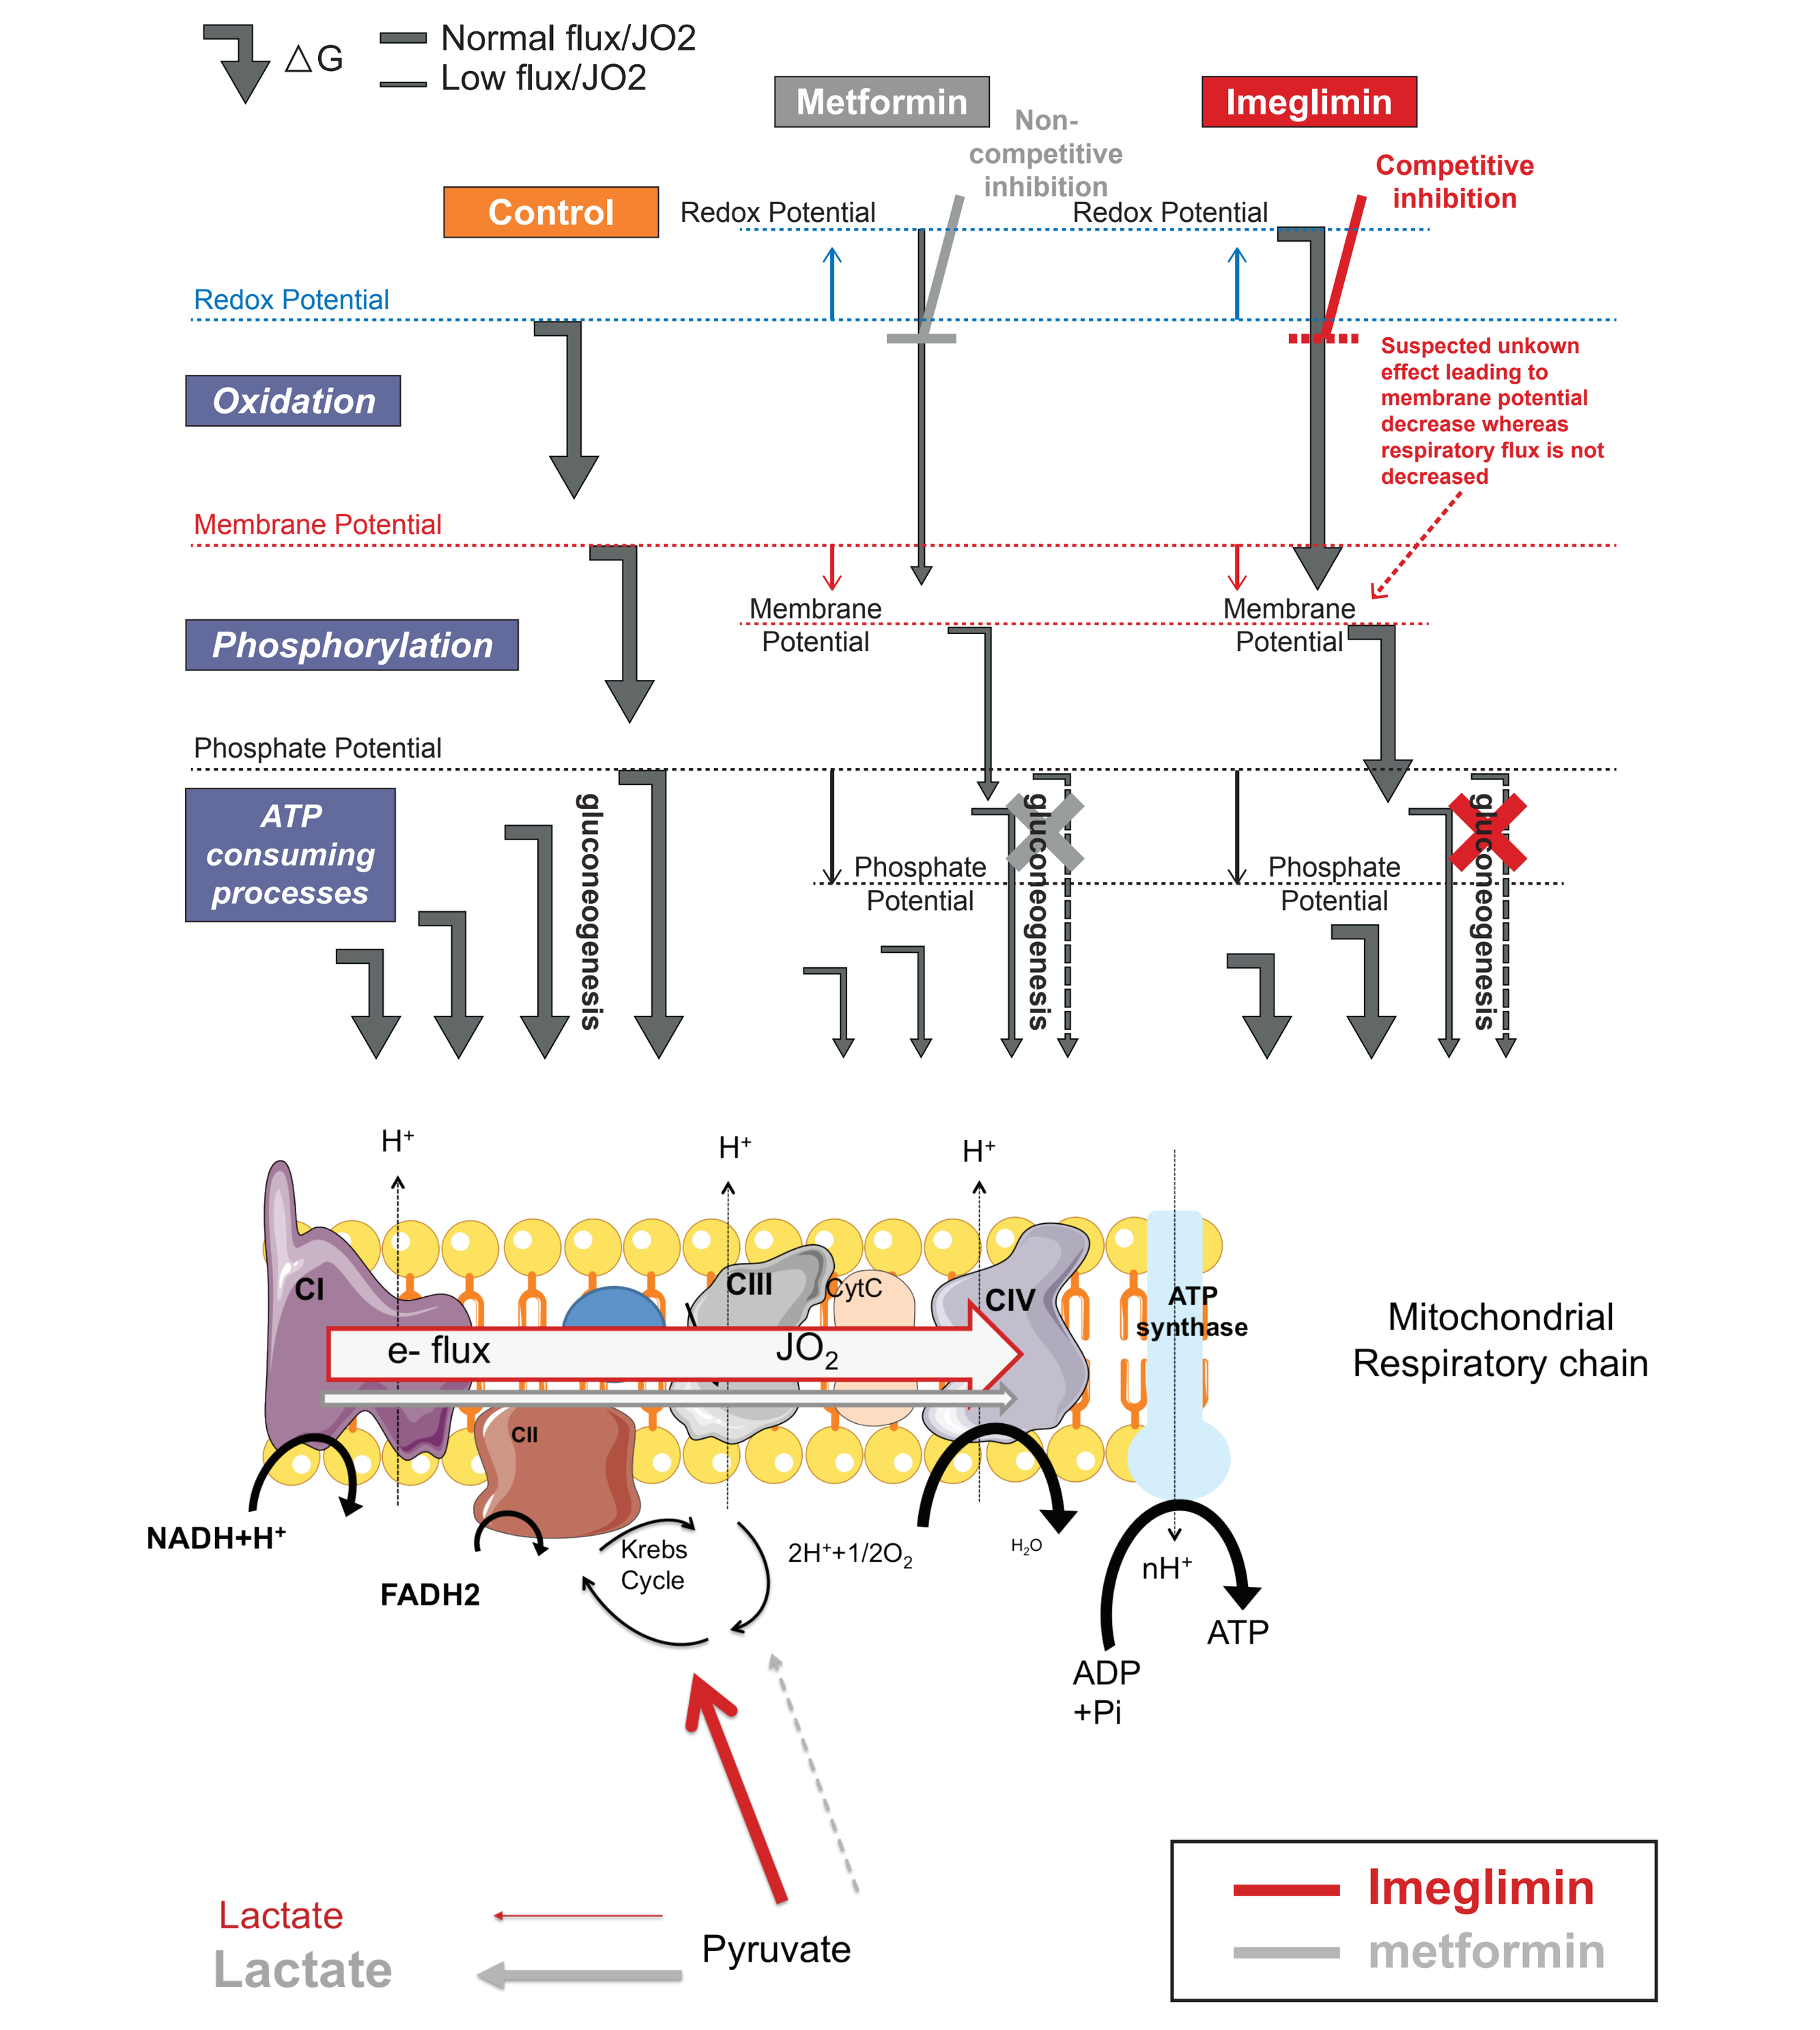
ΔG
Normal flux/JO2
Low flux/JO2
Metformin
Imeglimin
Non-competitive inhibition
Competitive inhibition
Control
Redox Potential
Redox Potential
Redox Potential
Oxidation
Suspected unkown effect leading to membrane potential decrease whereas respiratory flux is not decreased
Membrane Potential
Membrane Potential
Membrane Potential
Phosphorylation
Phosphate Potential
ATP consuming processes
gluconeogenesis
Phosphate Potential
Phosphate Potential
gluconeogenesis
gluconeogenesis
H+
H+
H+
CI
CII
CIII
CytC
CIV
ATP synthase
e- flux
JO_2
Mitochondrial Respiratory chain
NADH+H+
FADH2
Krebs Cycle
$2H^+ + 1/2O_2$
H_2O
nH+
ADP +Pi
ATP
Lactate
Lactate
Pyruvate
Imeglimin
metformin

References

1. Pirags V et al., Diabetes, Obesity and Metabolism;14:852-858.
2. Fouqueray P et al., Diabetes Care 2013;36(3):565–568.
3. Fouqueray P et al., Diabetes Care 2014;10:1935-5548.
4. Fouqueray P et al., J Diabetes Metab 2011;2:4.

U 1055 | Instituts thématiques Inserm Institut national de la santé et de la recherche médicale | CHU GRENOBLE | Université Joseph Fourier GRENOBLE | UNIVERSITÉ DE LYON | CarMeN LYON

Sophie Hallakou-Bozec PhD,
sophie.bozec@poxelpharma.com

poxel Excellence in achieving PoC