A Recombinant Antibody to EphA3 with Pro-Apoptotic and Enhanced ADCC Activity Against Various Hematologic Malignancies Shows Selective Inhibition of Colony Formation From Long-Term Culture-Initiating Cells (LTC-ICs) In Primary Leukemia Samples

Varghese Palath, PhD[1], Rohini Vekhande[1*], Mark Baer, PhD[1*], Wendy Ching, PhD[1*], Nenad Tomasevic, PhD[1*], David Martinez[1*], Christina Yi[1*], Swathi Bhaskar[1*], Kenneth Luehrsen, PhD[1*], Jason Williams[1*], John Leung[1*], Alan F. List, MD[2], Ling Zhang, MD[3], Martin Lackmann, PhD[4*], Andrew Boyd, MD, PhD[5], Andrew Scott, PhD[6*], Daniela Cilloni, MD, PhD[7*], Geoffrey Yarranton, PhD[1*] and Christopher Bebbington, DPhil.[1*]

[1]KaloBios Pharmaceuticals, Inc., South San Francisco, CA; [2]H. Lee Moffitt Cancer Center, Tampa, FL; [3]Department of Hematopathology and Clinical Medicine, H. Lee Moffitt Cancer Center, Tampa, FL; [4]Monash University, Clayton Victoria, Australia; [5]Queensland Inst. of Med. Rsch., Royal Brisbane Hospital, Brisbane, QLD, Australia; [6]Ludwig Institute of Cancer Research, Melbourne, Australia; [7]Dept. of Clinical and Biological Sciences, University of Turin, Orbassano, Italy

ABSTRACT

EphA3 is a receptor tyrosine kinase expressed at high level on the surface of some solid tumors and certain leukemias. KB004 is a high-affinity non-fucosylated, recombinant antibody developed by KaloBios derived from a mouse monoclonal anti-EphA3 antibody using Antibody Humaneering™ Technology. The antibody binds EphA3 from human and cynomolgus macaque with equivalent affinity (610 pM for human and 570 pM for cynomolgus) as determined by surface plasmon resonance (spr) analysis, making the latter a relevant toxicology species. KB004 has a direct pro-apoptotic effect on EphA3-positive cells and induces potent antibody-dependent cellular cytotoxicity (ADCC) activity mediated by CD16-expressing effector cells. The non-fucosylated antibody has enhanced affinity for both human and cynomolgus CD16a. The affinity of KB004 for cynomolgus CD16a (7.2 nM, determined by spr analysis) was comparable to the affinity for the high-affinity variant of CD16a from humans (6.6 nM), indicating that KB004 has similar potential for ADCC in cynomolgus macaques.

We first demonstrated cell-surface expression of EphA3 by flow cytometry in approximately 50% of primary human leukemia samples. Here we extend the analysis to multiple myeloma (MM) in which 3/5 patient samples show EphA3 expression. EphA3 expression was confirmed in a panel of chronic myeloid leukemia (CML) samples [n=17] and in acute myeloid leukemia (AML) [n=42] in which EphA3+ cells were identified in each French American British (FAB) classification sub-type. CD123-positive leukemia stem cells ($CD34^+$ $CD38^-$ $CD123^+$) also displayed surface expression of EphA3.

KB004 induced apoptosis in $EphA3^+$ primary cells from each of the diseases analyzed, including imatinib-sensitive and resistant CML, with no activity in EphA3-negative specimens, including normal $CD34^+$ bone marrow cells. KB004 inhibited myeloid leukemia colony formation (CFU-L) from primary AML samples in methylcellulose colony assays without affecting normal hematopoietic colony recovery and also showed dose-dependent inhibition of long-term culture-initiating cells (LTC-IC) from AML stem cells. EphA3 expression was not detected on primary human $CD34^+$ bone marrow cells from healthy individuals.

These data support the development of an anti-EphA3 antibody for the treatment of hematologic malignancies. The selective activity against leukemic stem cells supports a novel strategy for therapeutic targeting of leukemia-initiating cells.

INTRODUCTION

EphA3 was originally identified as an oncofetal antigen on a pre-B-cell leukemic cell line (LK63) (Boyd *et al* 1992; Wicks *et al* 1992) using a monoclonal antibody IIIA4. The antibody has been shown to recognize the highly conserved N-terminal domain of EphA3 adjacent to the ligand binding site but does not interfere with binding of the natural ligand, ephrin-A5 (Smith *et al* 1994). KB004 is a high-affinity recombinant human IgG1 antibody, with the binding specificity of IIIA4, derived by Antibody Humaneering™ Technology. This technology replaces non-human V-segments with sequences from a human V-region library such that the V-regions of the resulting antibody have high sequence identity with human germ-line sequence and are expected to show low immunogenicity in humans.

The antibody is produced from a CHO cell line deficient in α1,6 fucosyl transferase to generate antibody lacking fucose in its carbohydrate structure. Non-fucosylated antibodies have been shown to have enhanced ADCC activity mediated by CD16a (Mori *et al.* 2007).

RESULTS

Table 1: KB004 Fab Binds to Human and Cynomolgus EphA3 with Equivalent Affinity

Analyte	k_a ($M^{-1}s^{-1}$)	k_d (s^{-1})	K_D (pM)
Human EphA3-Fc	2.7×10^6	1.6×10^{-3}	610
Cynomolgus EphA3-Fc	3.0×10^6	1.7×10^{-3}	570

Binding of KB004 Fab to immobilized human or cynomolgus EphA3-Fc fusion protein was analyzed by surface plasmon resonance analysis (Biacore 3000) and global fit analysis. k_a = association constant; k_d = dissociation constant; K_D = affinity.

Figure 1: EphA3 protein is expressed on the cell surface in a proportion of primary samples from myeloid leukemias, myelodysplasias (MDS), myeloproliferative neoplasms (MPN) and multiple myeloma (MM)

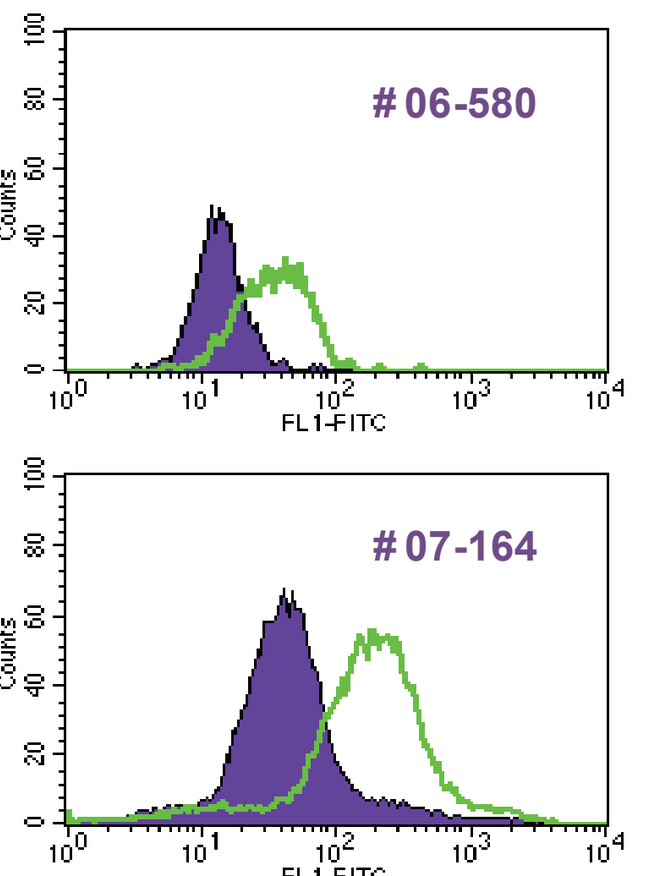
#06-580
#07-164
Counts
FL1-FITC

Flow-cytometry screen

Tumor type	Number of samples tested	EphA3 positive samples	Samples positive for EphA3 (%)
AML	42	25	59
CML	12	4	33
CML (IR)	5	4	80
MPN	6	3	50
MDS	16	7	44
MM	5	3	60

A panel of previously cryopreserved bone marrow or peripheral blood samples were tested for EphA3 expresssion by flow cytometry. Samples were scored positive if >5% of cells showed anti-EphA3 binding above the background due to an isotype control antibody. AML = acute myeloid leukemia; CML = chronic myeloid leukemia; MDS = myelodysplastic syndromes; MPN = myeloproliferative neoplasms; MM = multiple myeloma. CML patients were pre-treatment or undergoing therapy. IR = imatinib-resistant samples. Histograms for 2 representative AML patients are shown. Binding of anti-Epha3 antibody (green) is compared with human IgG1 isotype control (blue). Bound antibody was detected using anti-human IgG-FITC conjugate and fluorescence intensity was measured (FL1-FITC) using a FACSCaliber instrument.

Figure 2: Anti-EphA3 antibody binds to AML bone marrow $CD34^+$ cells and cells with a leukemic stem cell phenotype ($CD34^+$ $CD38^-$ $CD123^+$)

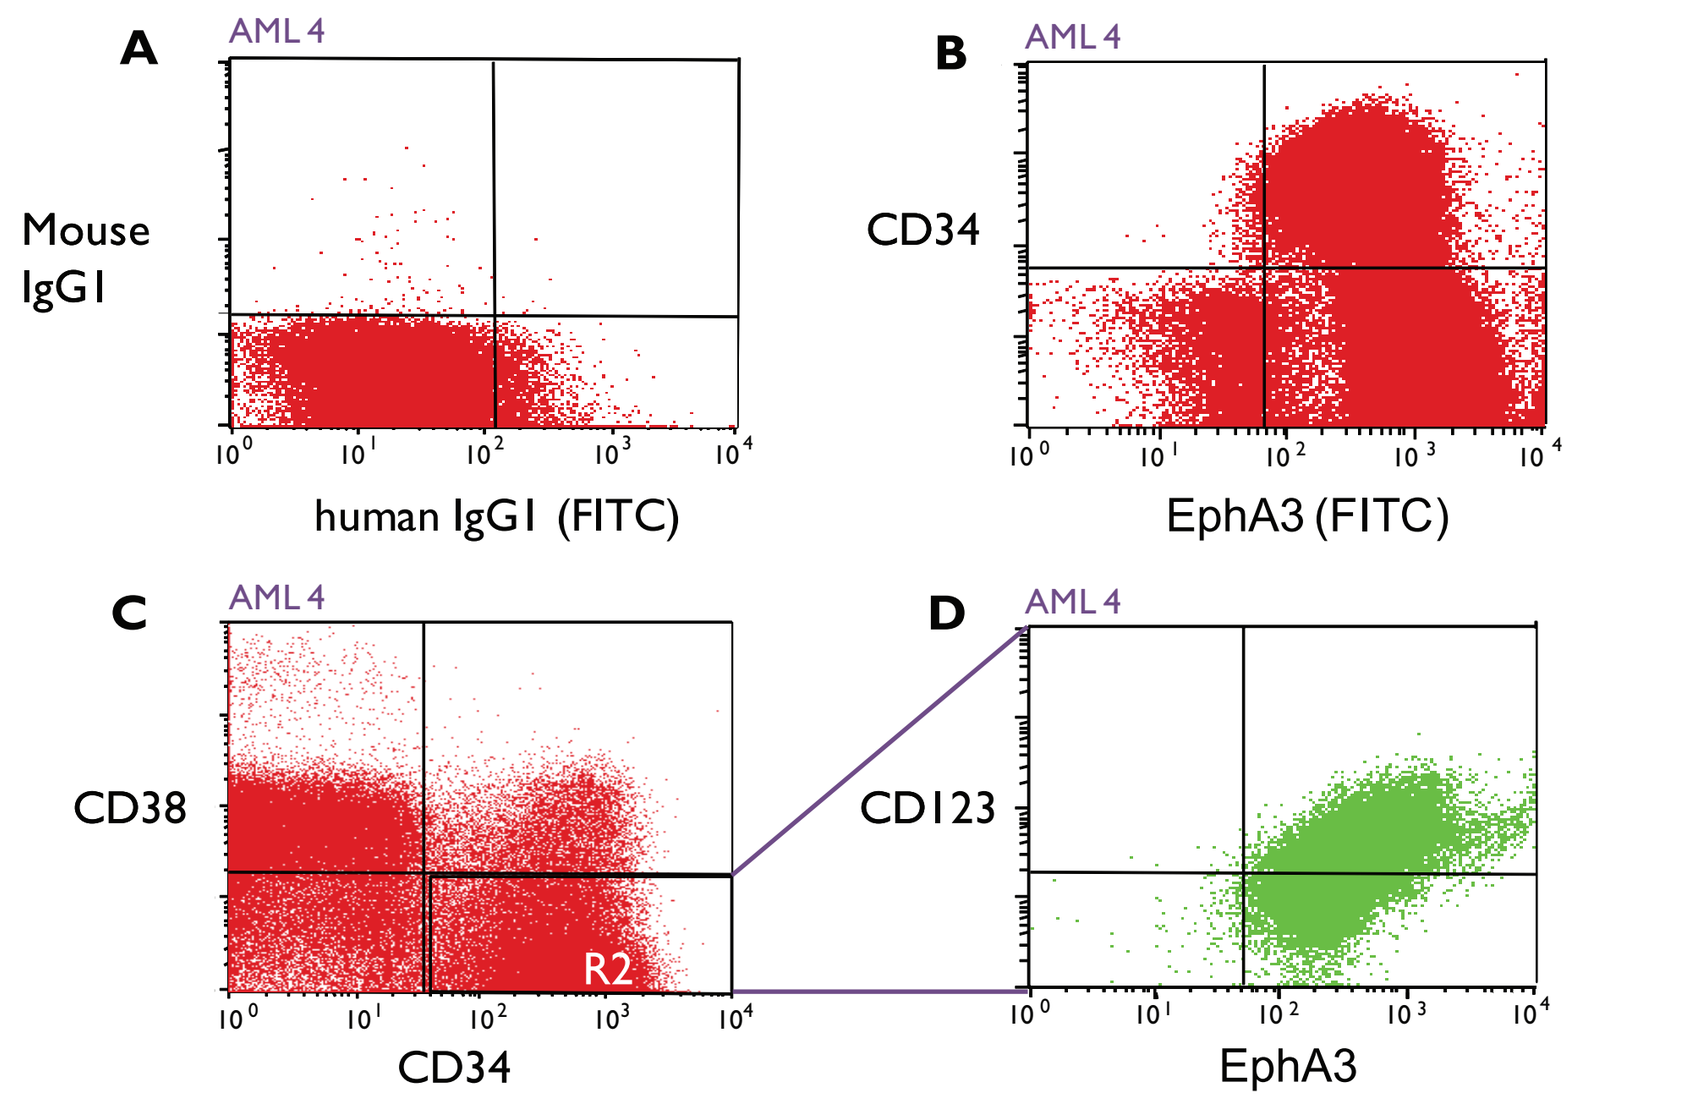
A
B
C
D
AML 4
Mouse IgG1
human IgG1 (FITC)
CD34
EphA3 (FITC)
CD38
CD34
R2
CD123
EphA3

AML primary bone marrow cells (Sample 08111462) were stained with: Humaneered anti-EphA3 antibody or IgG1 control and FITC-conjugated anti-human IgG; PE-conjugated anti-CD34; PEcy5-conjugated anti-CD38; and APC-conjugated anti-CD123 antibodies for flow cytometry analysis (50, 000 events per sample). (A) isotype control gating for CD34 and EphA3 analysis. (B) Sample stained with anti-EphA3 and anti-CD34. (C) Sample stained for CD34 and CD38 (R2 represents $CD34^+$ $CD38^-$ cells). (D) Identification of EphA3 and CD123 expression on $CD34^+$ $CD38^-$ cells (R2 gate).

Figure 3. EphA3 is not detectable on normal human bone marrow $CD34^+$ cells.

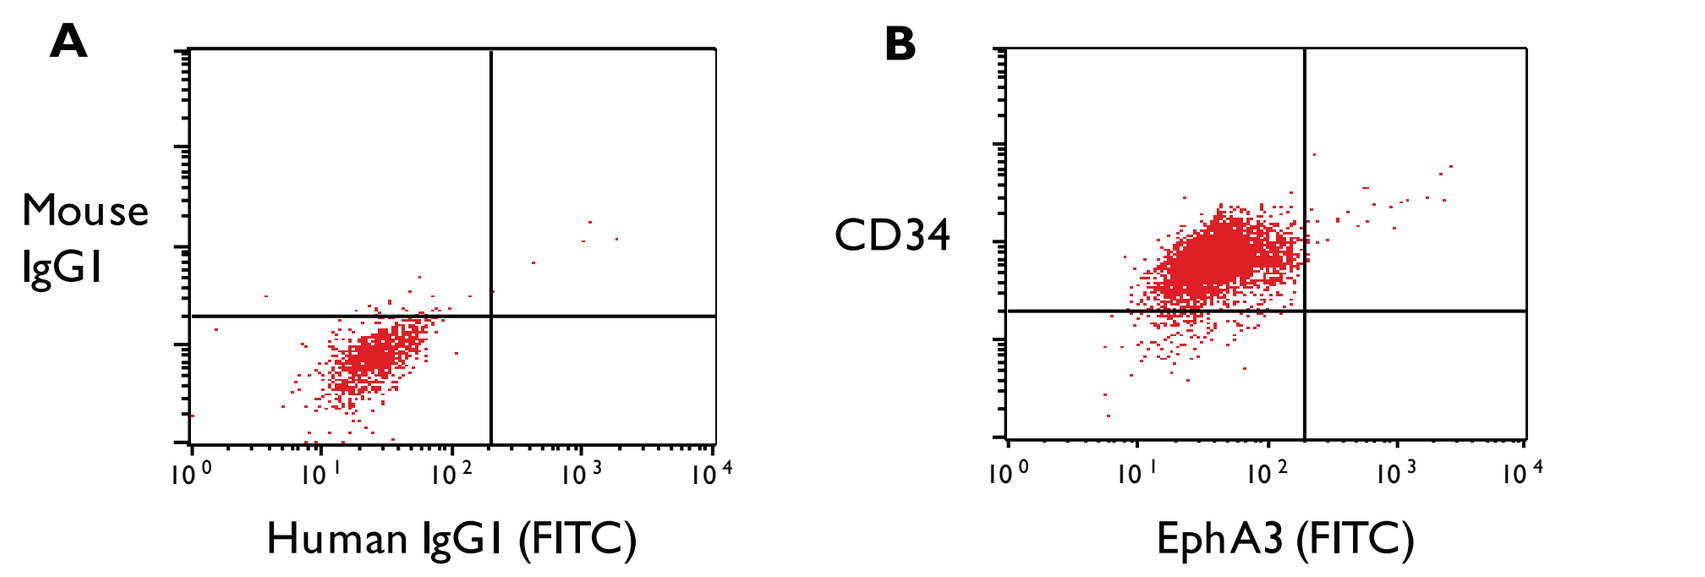
A
B
Mouse IgG1
Human IgG1 (FITC)
CD34
EphA3 (FITC)

Normal human bone marrow $CD34^+$ cells were stained with Humaneered anti-EphA3 antibody or control IgG1 and FITC-conjugated anti-human IgG and PE-conjugated anti-CD34 antibody and analyzed by flow cytometry (5000 events per sample). (A) isotype control gating for $CD34^+$ and EphA3 cell analysis. (B) staining with anti-EphA3 and anti-CD34.

Figure 4. Humaneered anti-EphA3 antibody induces apoptosis in $EphA3^+$ primary leukemia cells.

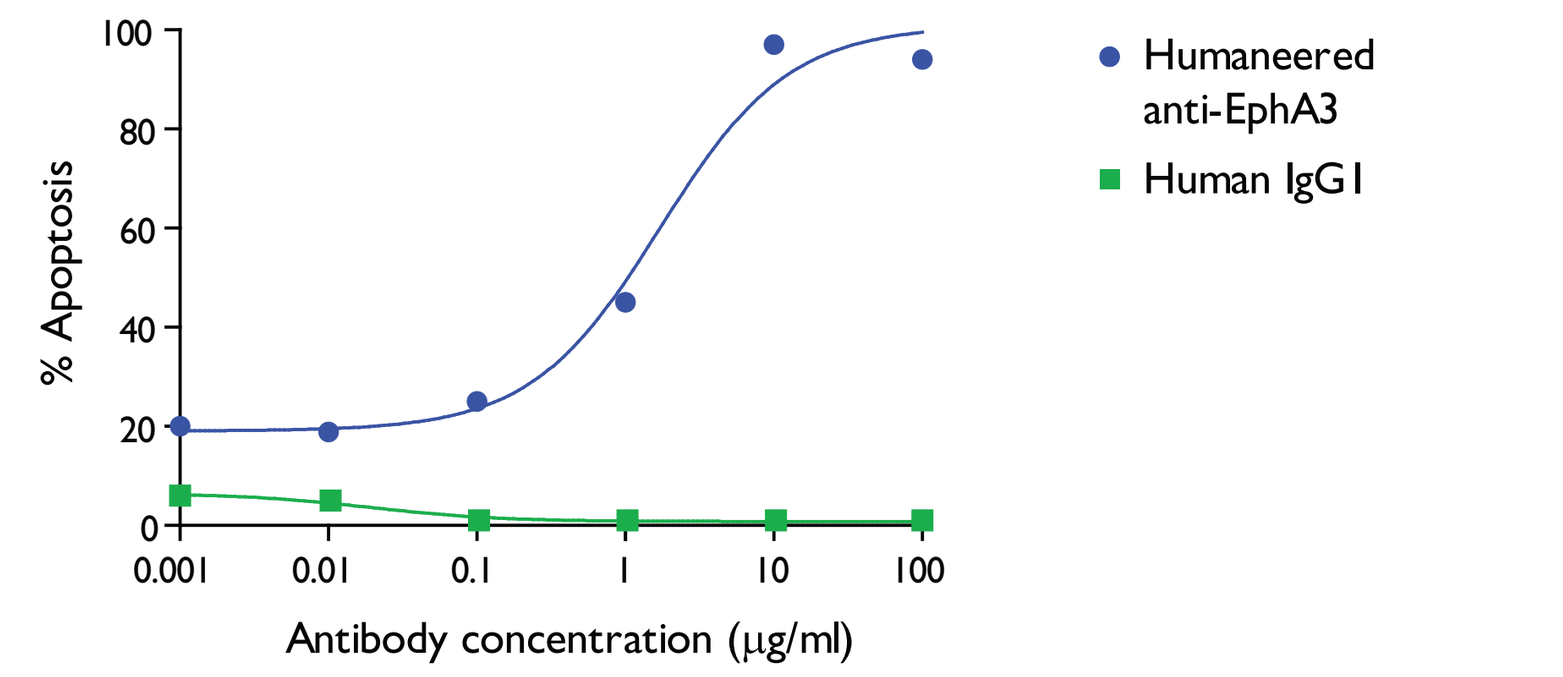
Humaneered anti-EphA3
Human IgG1
% Apoptosis
Antibody concentration (μg/ml)

Humaneered anti-EphA3 (fucosylated) or isotype control antibody was added to CML Bone Marrow cells (Sample 7179) for 24 h. Apoptosis was determined by Annexin V and Propidium Iodide staining.

Figure 5. Fucosylated Humaneered anti-EphA3 antibody induces ADCC. Non-fucosylated antibody (KB004) has enhanced ADCC activity.

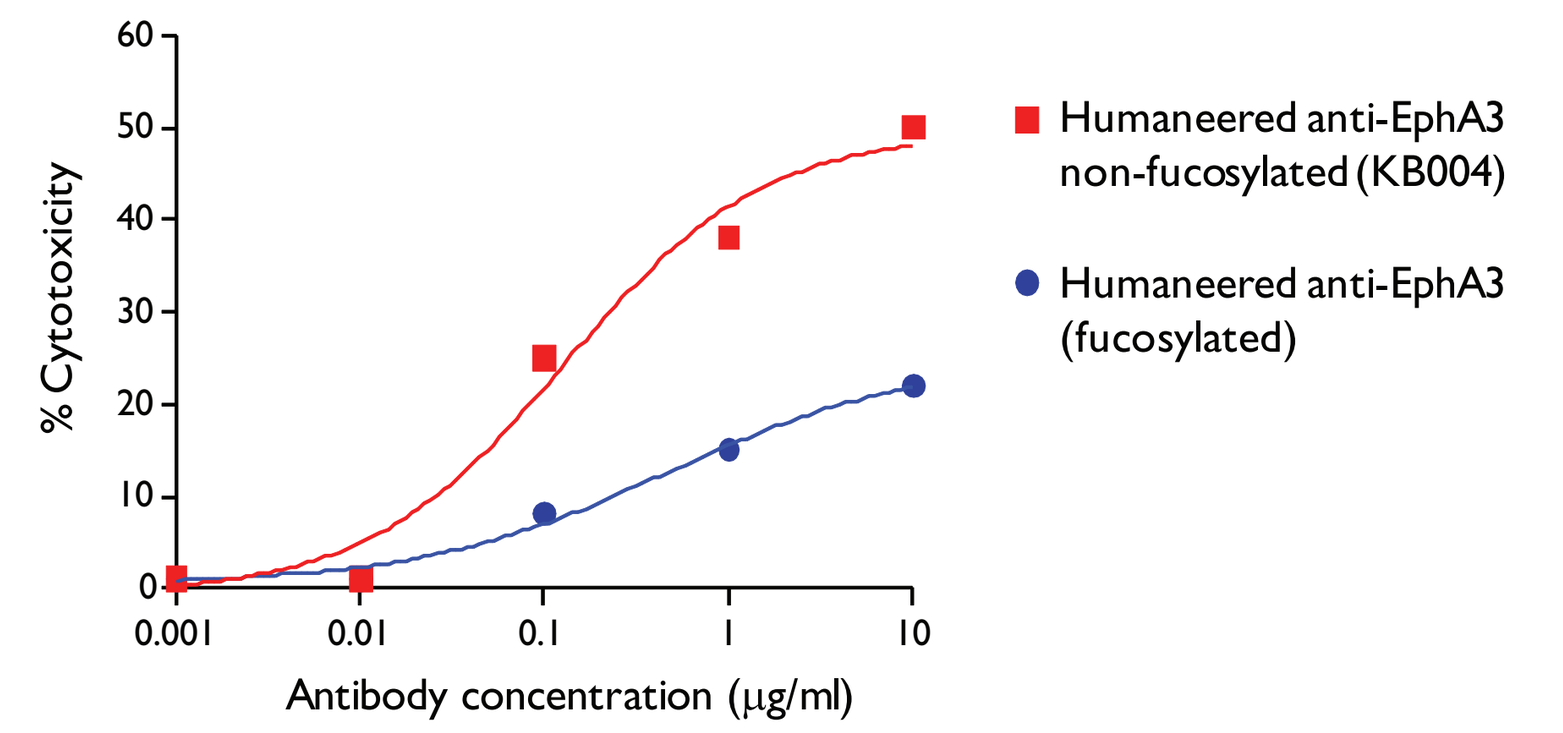
Humaneered anti-EphA3 non-fucosylated (KB004)
Humaneered anti-EphA3 (fucosylated)
% Cytotoxicity
Antibody concentration (μg/ml)

KB004 or its fucosylated counterpart was added to primary patient sample target cells (RBH020) together with PBMC effector cells for 16 hours at an effector: target cell ratio of 100:1.

Table 2: Non-fucosylated KB004 has a higher affinity for recombinant CD16a than does its fucosylated counterpart

Analyte	k_a ($M^{-1}s^{-1}$)	k_d (s^{-1})	K_D (nM)
KB004 (non-fucosylated)	8.2×10^5	4.9×10^{-3}	6
KB004 (fucosylated)	7.6×10^4	5.1×10^{-3}	66

Binding of fucosylated and non-fucosylated antibody forms to immobilized CD16a (high-affinity variant 158V) was analyzed by surface plasmon resonance analysis (Biacore 3000) and global fit analysis assuming a 1:1 binding interaction.

Figure 6: KB004 inhibits growth of leukemic colonies from AML bone marrow samples cultured in methylcellulose

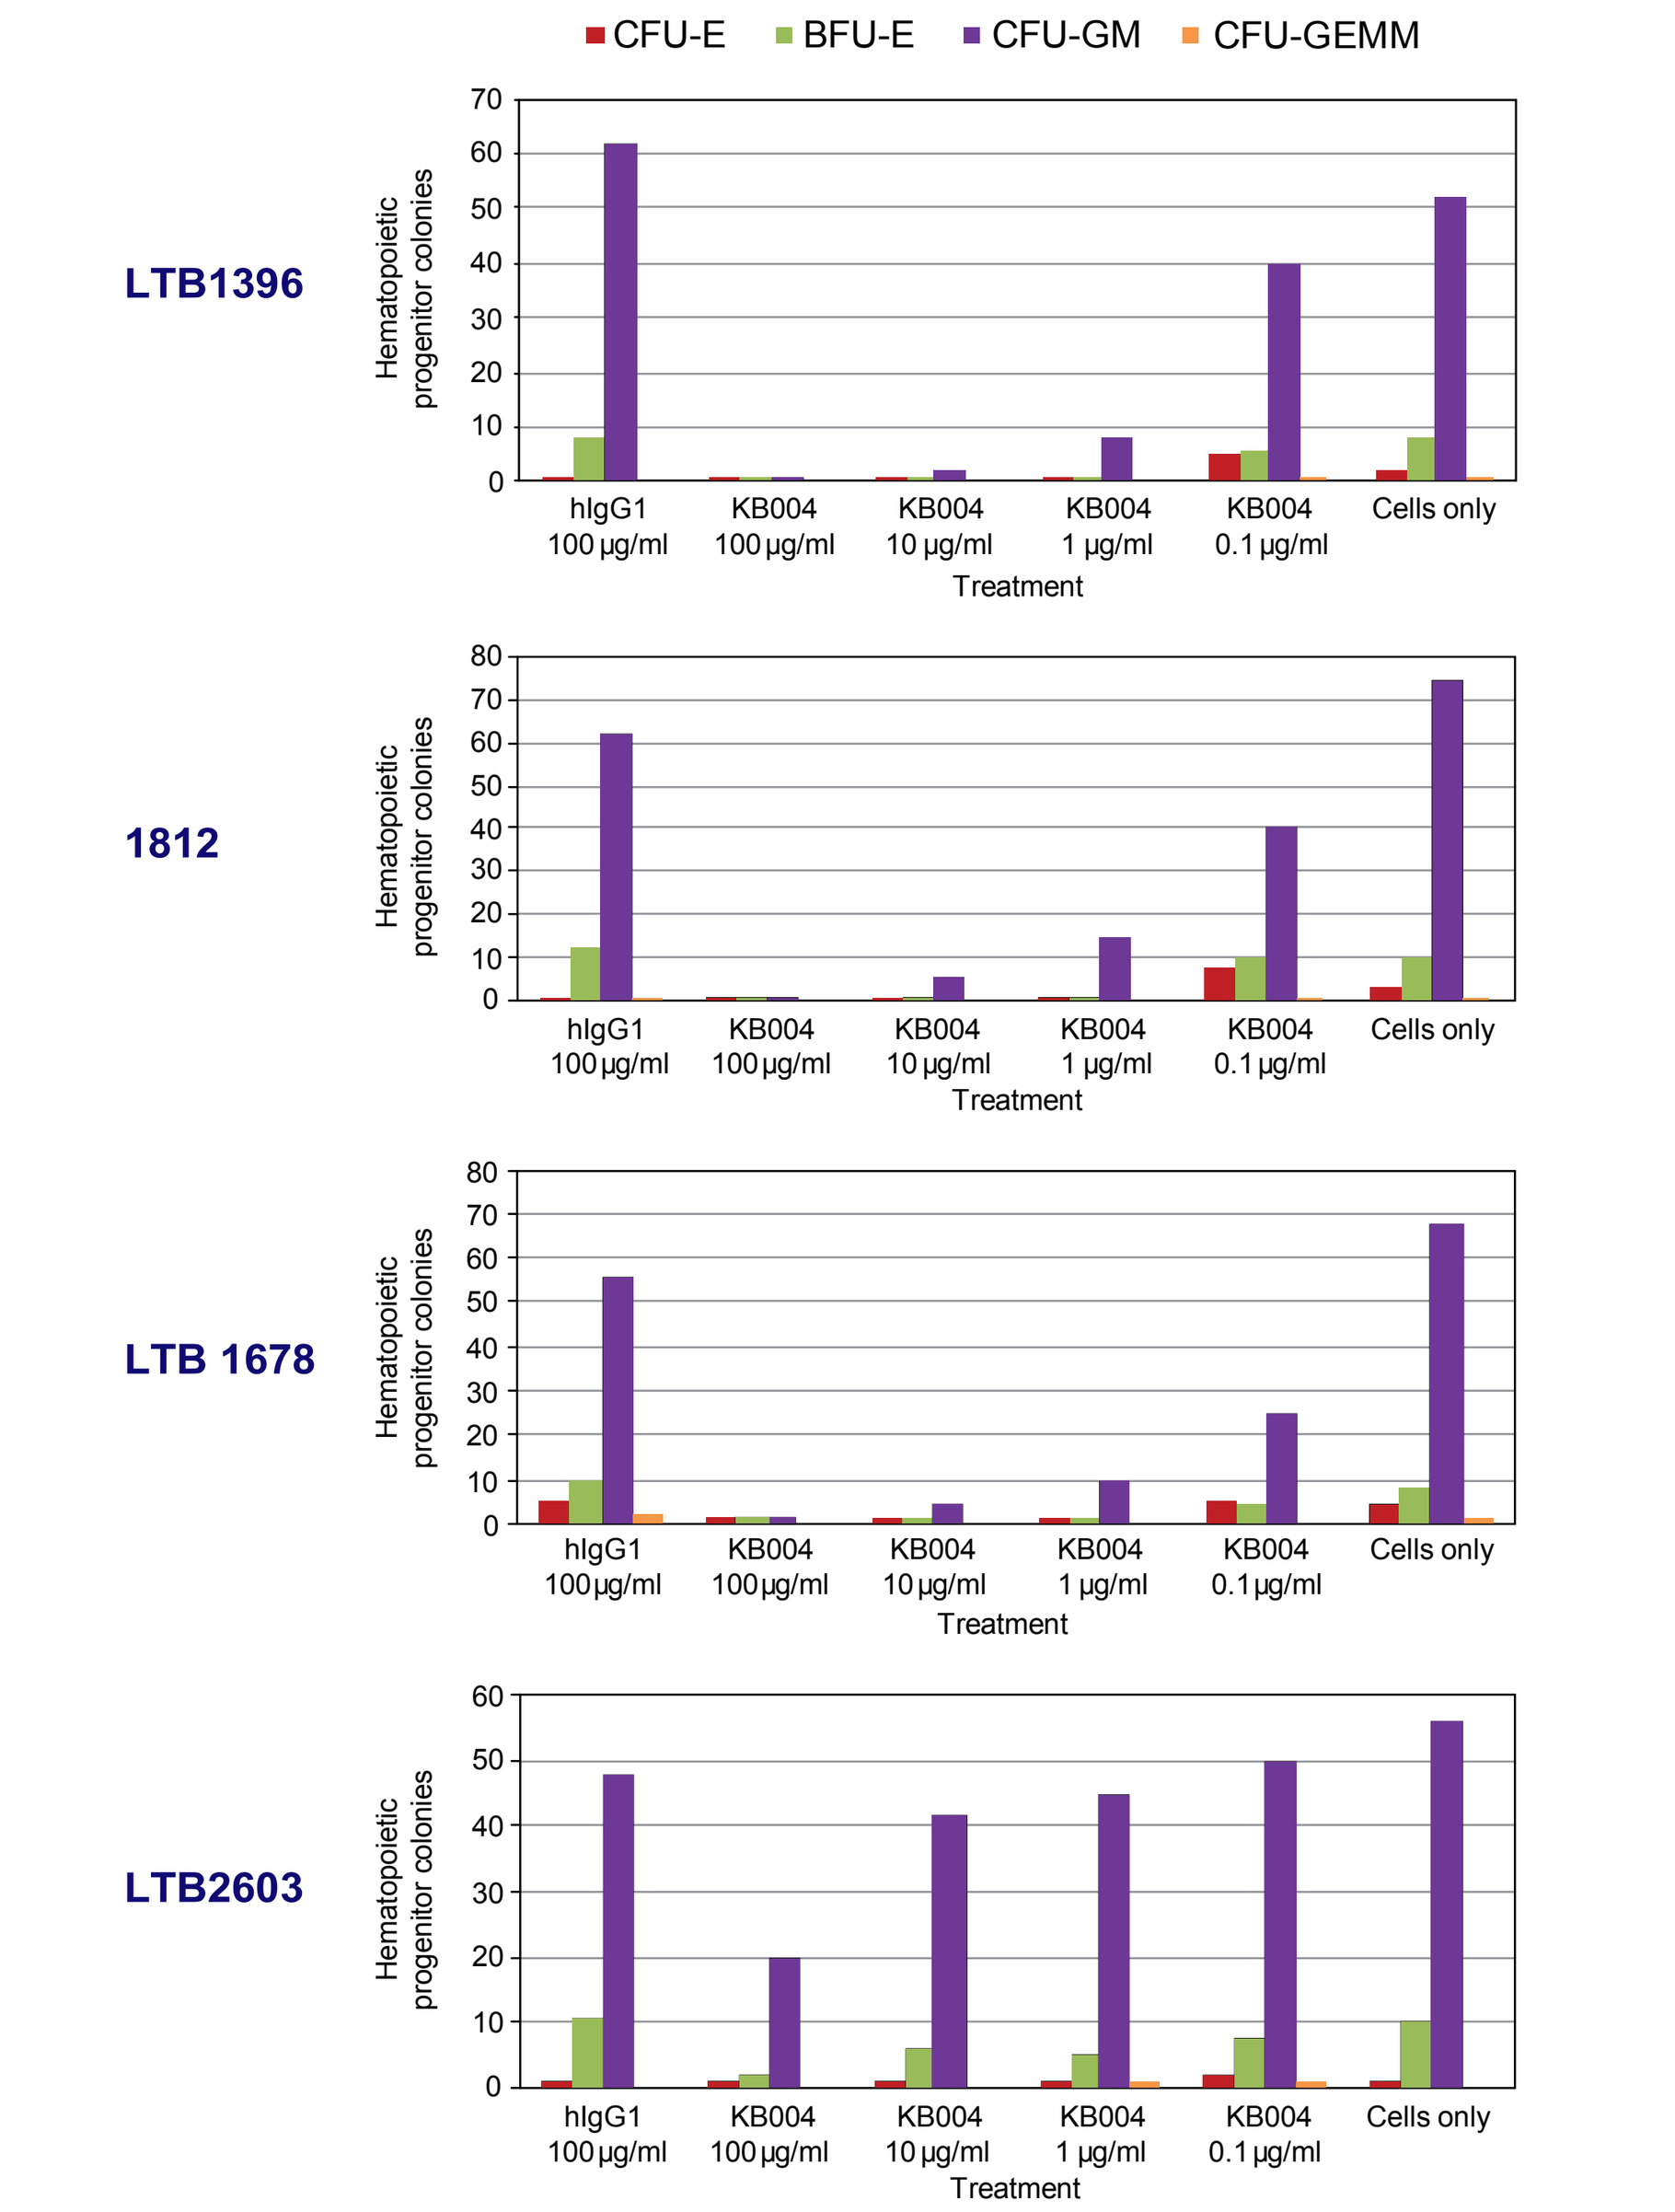
CFU-E
BFU-E
CFU-GM
CFU-GEMM
LTB1396
1812
LTB 1678
LTB2603
Hematopoietic progenitor colonies
hIgG1 100 μg/ml
KB004 100 μg/ml
KB004 10 μg/ml
KB004 1 μg/ml
KB004 0.1 μg/ml
Cells only
Treatment

AML bone marrow cells (1.0×10^5 cells/mL) from 4 patients were plated in Methocult H4434 methylcellulose containing 10 ng/mL GM CSF, 10 ng/mL IL-3, 50 ng/mL SCF, and 3 U/mL EPO with or without KB004 or control antibody in 35-mm culture dishes. Hematopoietic progenitor colonies of various morphologies were counted after 14 days. CFU-E = erythroid colony-forming units; BFU-E = erythroid burst-forming units; CFU GM = granulocyte-macrophage colony-forming units; CFU GEMM = multipotential granulocyte, erythrocyte, monocyte, megakaryocyte colony-forming units; EPO = erythropoietin; GM-CSF = granulocyte-macrophage colony stimulating factor; SCF = stem cell factor.

Figure 7: KB004 antibody shows dose-dependent Inhibition of long-term culture-initiating cells (LTC-IC) from AML stem cells.

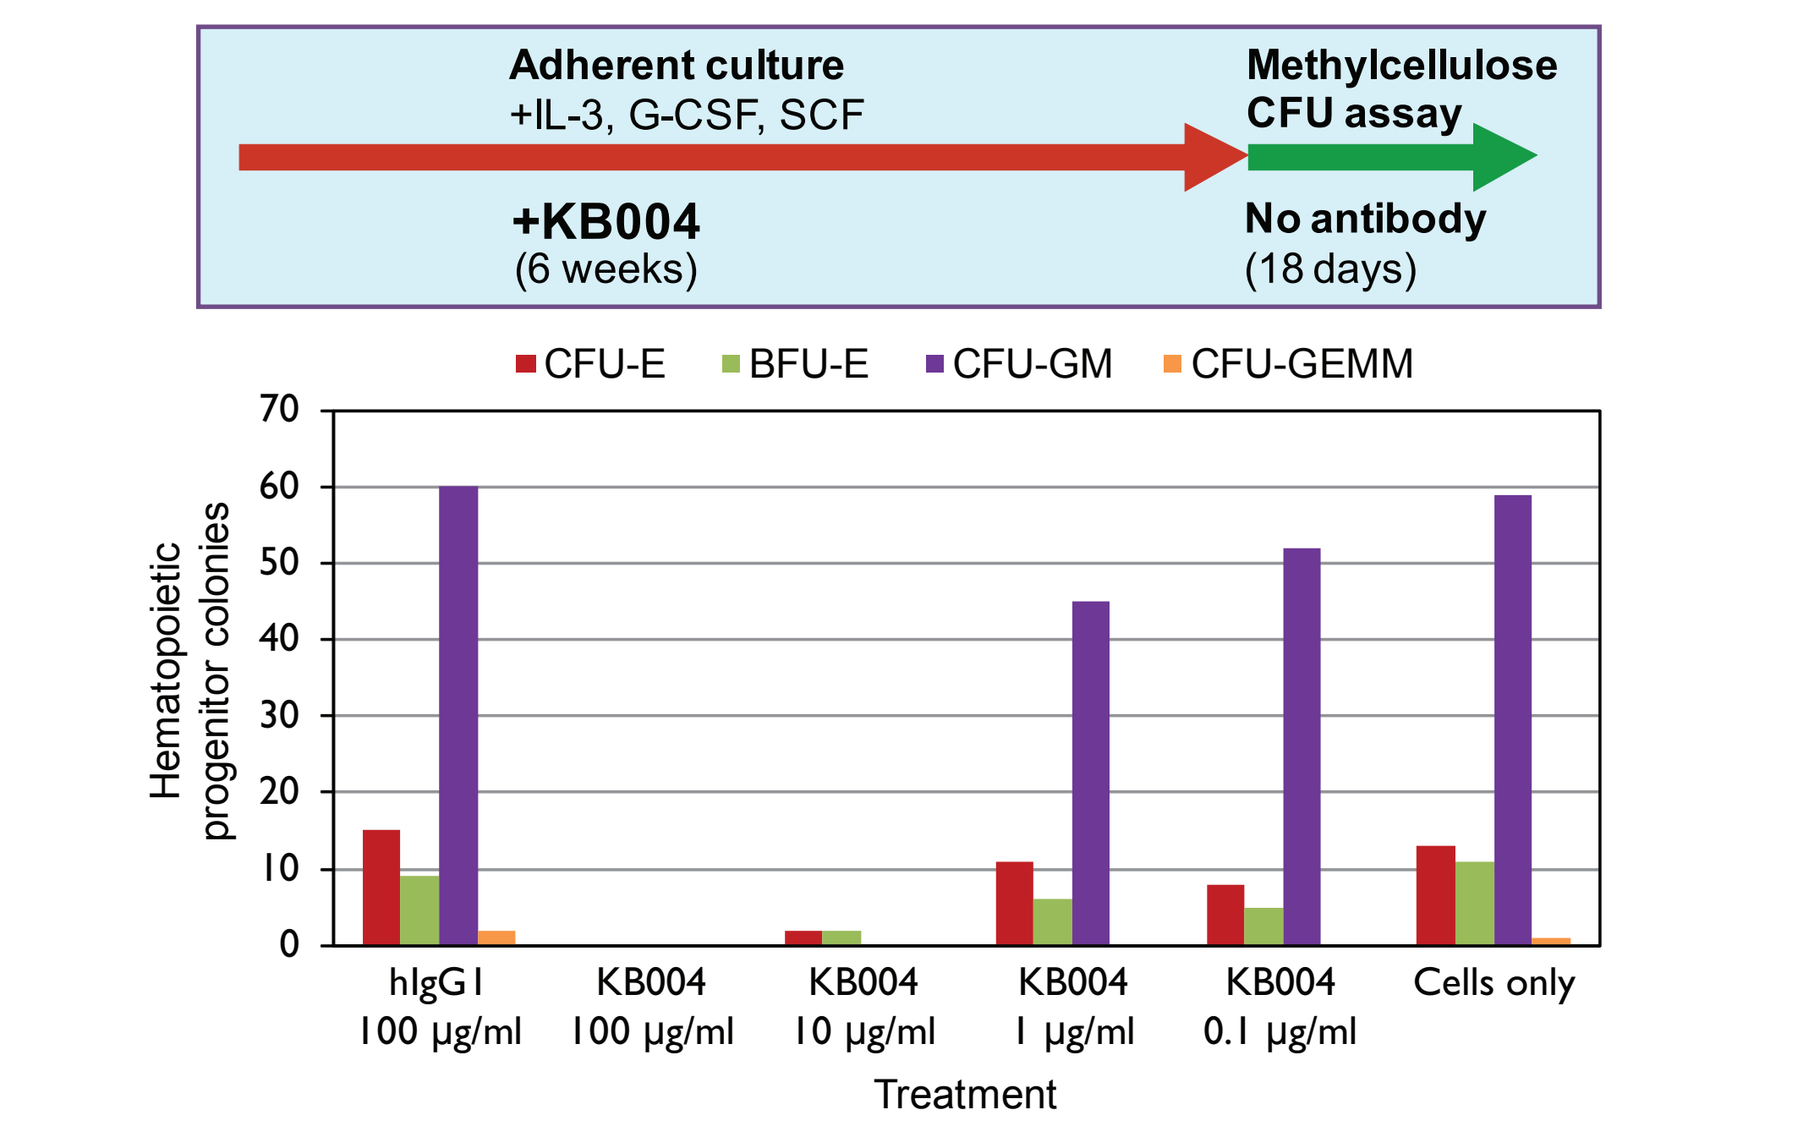
Adherent culture
+IL-3, G-CSF, SCF
+KB004
(6 weeks)
Methylcellulose CFU assay
No antibody
(18 days)
CFU-E
BFU-E
CFU-GM
CFU-GEMM
Hematopoietic progenitor colonies
hIgG1 100 μg/ml
KB004 100 μg/ml
KB004 10 μg/ml
KB004 1 μg/ml
KB004 0.1 μg/ml
Cells only
Treatment

Primary M2 AML cells (sample PAH019 from peripheral blood) were cultured on mitomycin-treated transfected fibroblast lines secreting IL-3, SCF and G-CSF for 6 weeks in MyeloCult medium with 10^{-6}M hydrocortisone in the presence of KB004 or control antibody. Adherent tumor stem cells were then transferred to methylcellulose (Methocult) containing, IL-3, IL-6, SCF, GM-CSF, G-CSF and EPO without KB004 to determine surviving colony-forming units.

Figure 8: KB004 antibody does not affect growth of hematopoietic cell colonies from normal human bone marrow $CD34^+$ cells.

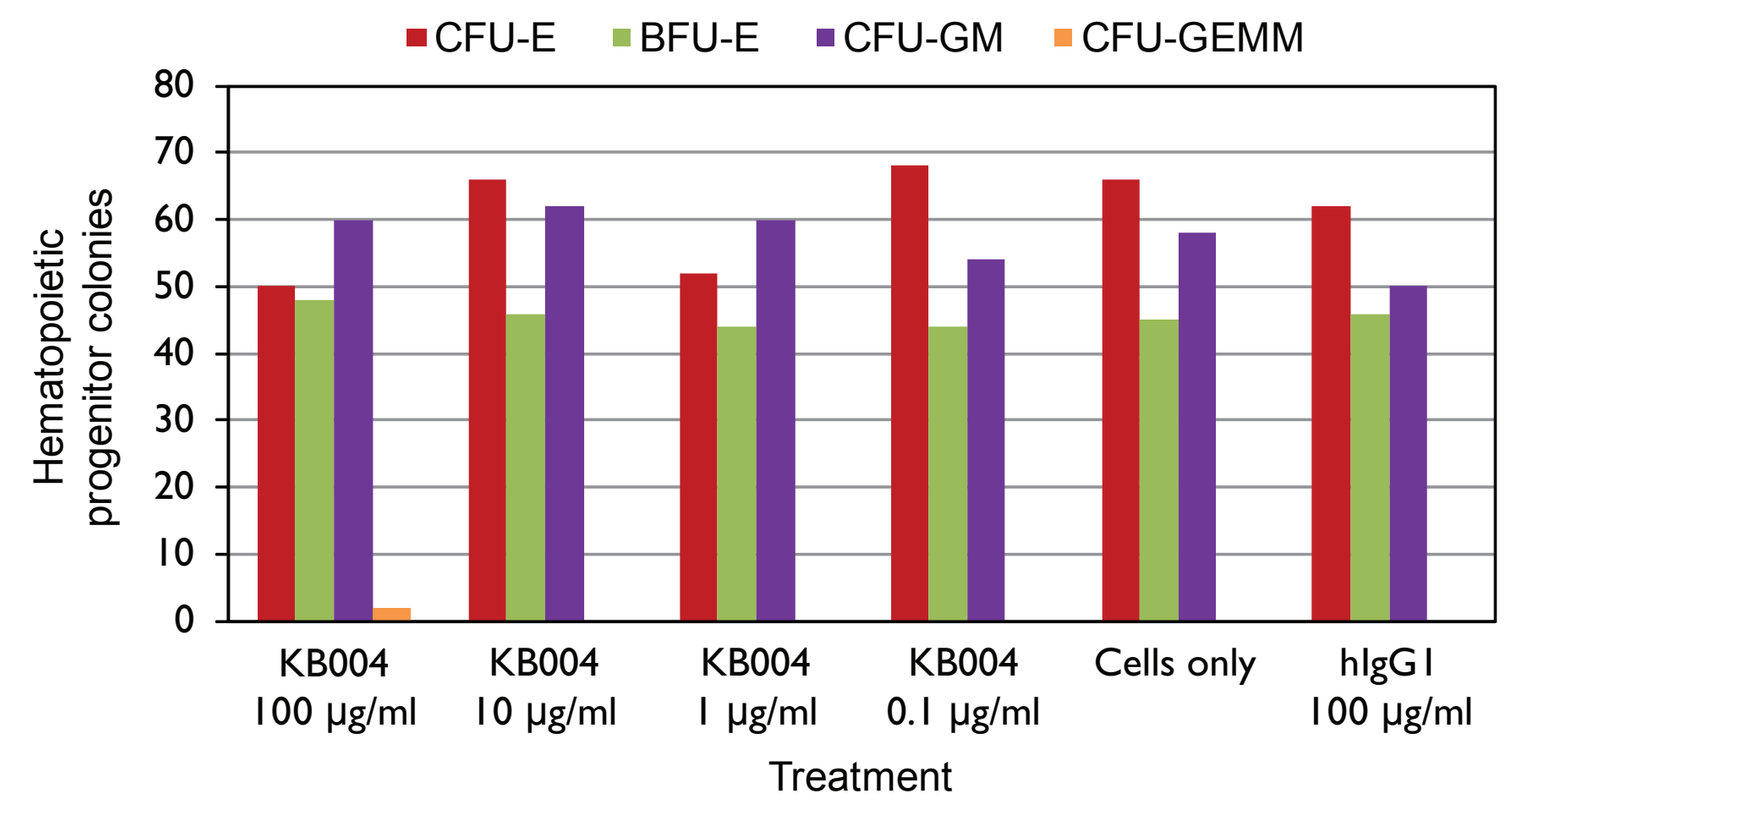
CFU-E
BFU-E
CFU-GM
CFU-GEMM
Hematopoietic progenitor colonies
KB004 100 μg/ml
KB004 10 μg/ml
KB004 1 μg/ml
KB004 0.1 μg/ml
Cells only
hIgG1 100 μg/ml
Treatment

Normal bone marrow $CD34^+$ cells (500 cells/ ml) were plated in MethoCult with or without KB004 or control IgG1 antibody in 35 mm culture dishes. Hematopoietic progenitor cell colonies (BFU-E, CFU-E, CFU-GM and CFU-GEMM) were counted after 14 days.

Figure 9: KB004 antibody does not affect the growth of long-term culture-initiating cells (LTC-IC) from normal human bone marrow $CD34^+$ cells.

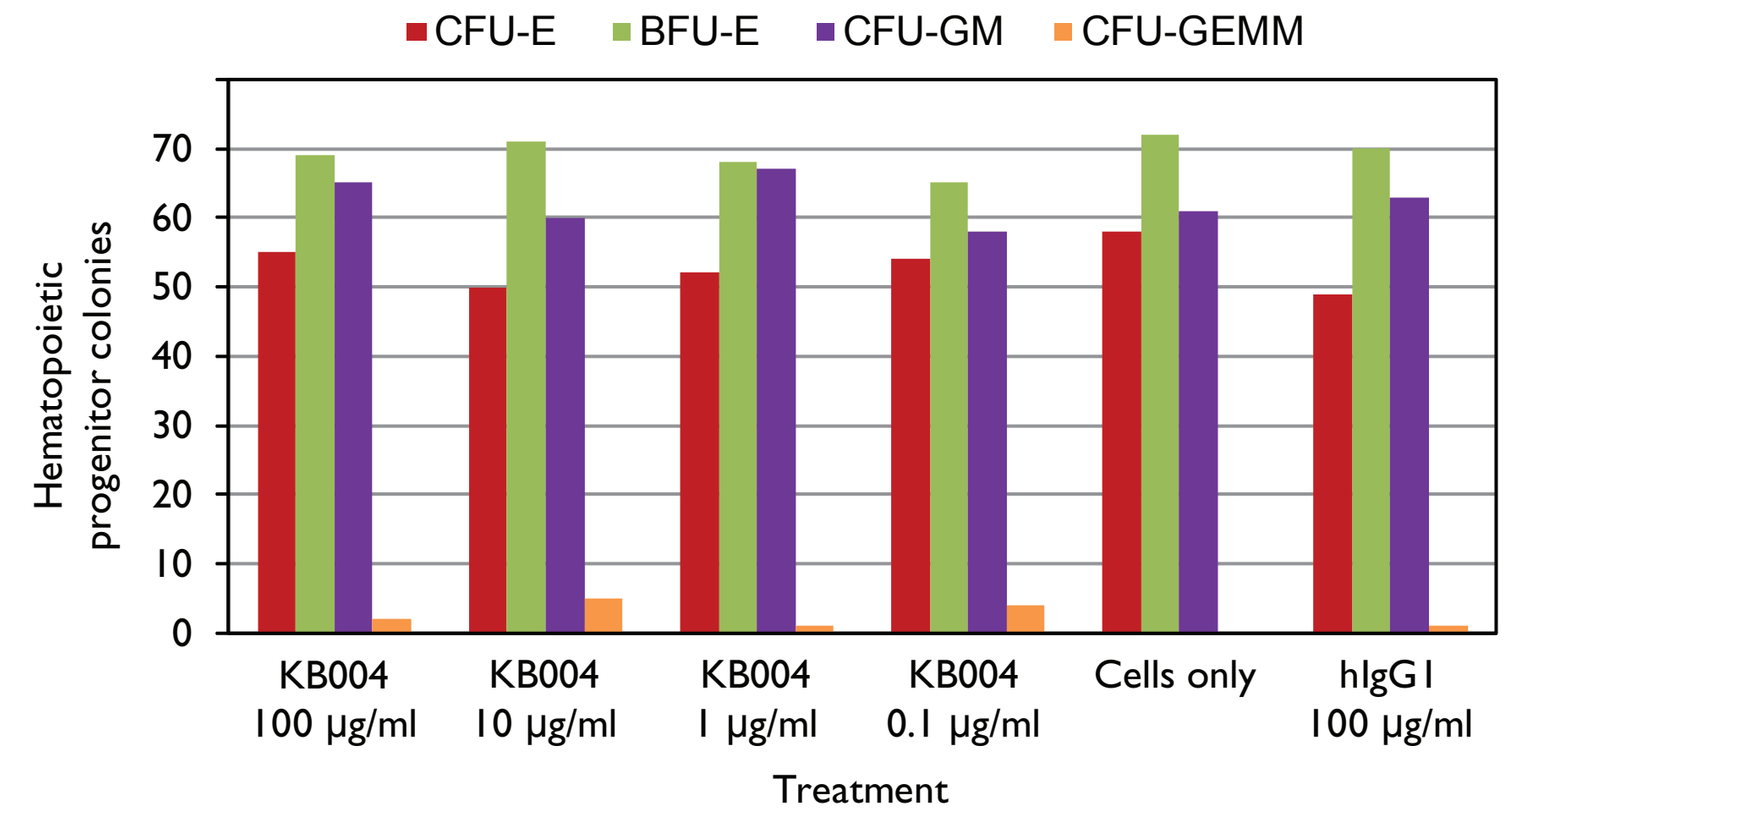
CFU-E
BFU-E
CFU-GM
CFU-GEMM
Hematopoietic progenitor colonies
KB004 100 μg/ml
KB004 10 μg/ml
KB004 1 μg/ml
KB004 0.1 μg/ml
Cells only
hIgG1 100 μg/ml
Treatment

Purified $CD34^+$ cells from normal bone marrow were cultured on mitomycin-treated transfected fibroblast lines secreting IL-3, SCF and G-CSF for 6 weeks in MyeloCult medium with 10^{-6}M hydrocortisone in the presence of KB004 or control antibody. Adherent stem cells were then transferred to methylcellulose (Methocult) containing, IL-3, IL-6, SCF, GM-CSF, G-CSF and EPO without KB004 to determine surviving colony-forming units.

Table 3: KB004 induces apoptosis in AML Cells expressing EphA3 on the cell surface and mediates ADCC activity against $EphA3^+$ cells

Sample	AML Type	$EphA3^+$ (%)	KB004 Activity: ADCC (%)	KB004 Activity: Apoptosis (%)	$EphA3^+$ (% of $CD34^+$)	$EphA3^+$ (% of $CD34^+$ $CD38^-$)	LSC ($CD123^+$ $CD38^-$ $CD34^+$): LSC (%)	LSC ($CD123^+$ $CD38^-$ $CD34^+$): $EphA3^+$ (% of LSC)
LTB1678	M4	99	95	100	100	100	4	100
LTB237	M0	98	90	99	100	100	0	0
LTB1812	M4[a]	98	80	100	100	100	0	0
LTB383	M1[a]	96	94	98	100	100	0	0
08111462	M5b	88	95	79	100	100	25	100
07-164	M2	84	47	50	100	100	N/D	N/D
LTB1396	M5a	74	75	80	73	70	0	0
LTB2472	M5b	66	70	68	74	100	0	0
LTB1222	ex RAEB	65	N/D	N/D	95	100	14	100
LTB1265	M6	56	60	55	100	N/A	0	0
157260	M2	55	72	79	64	0	0	0
PAH019	M2	55	65	75	99	100	0	0
06-580	M3	51	86	86	98	N/A	0	0
RBH020	M1	41	50	48	92	100	0	0
148954	M5b	31	40	45	36	N/A	0	0
LTB2603	M4	27	25	25	78	63	0	0
161244	M3	15	22	20	N/A	N/A	0	0
06229	M7	21	20	22	100	99	12	99
160600	M2	16	20	15	12	100	10	100
LTB2490	M5b[a]	14	20	15	22	100	0	0
LTB1772	M5b	8	5	7.5	55	55	0	0
07-75	N/A	0	0	0	0	0	0	0

Primary AML samples were stained with anti-EphA3 antibody, PE-conjugated anti-CD34, PEcy5-conjugated anti-CD38 and APC-conjugated anti-CD123 for flow cytometry analysis. For ADCC assays, 10 μg/mL KB004 was added to primary patient sample target cells together with PBMC effector cells from healthy volunteers at an effector-to-target cell ratio of 100:1 for 16 hours. For apoptosis assays, 10 μg/mL KB004 was added to target cells for 24 hours. N/A = Not applicable. N/D = Not determined. In vitro killing of $EphA3^+$ cells by ADCC and apoptosis correlates with the number of cells expressing detectable levels of EphA3. Samples are from bone marrow aspirates except PAH019 which is from peripheral blood.

Table 4: KB004 Induces apoptosis in CML Cells expressing EphA3 on the cell surface and mediates ADCC activity against $EphA3^+$ cells.

Sample	Imatinib status	$EphA3^+$ (%)	$CD34^+$ (%)	$EphA3^+$ (% of $CD34^+$)	$EphA3^+$ (% of $CD34^+CD38^-$)	KB004–mediated ADCC (% total cells)	KB004–mediated Apoptosis (% total cells)	Imatinib mediated Apoptosis (%)
BM 7179(I)	unknown	100	23	100	N/A	80	85	N/D
PB 640 (I)	unknown	71	41	100	N/A	75	70	N/D
BM 500 (I)	unknown	59	0	0	N/A	70	70	N/D
BM 100001288	Pre-tx	95	0	N/A	N/A	95	82	58
PB F7040A43	active	0	25	0	0	0	0	55
BM P050310(I)	refractory	100	100	100	100	92	100	0
BM P051710(I)	refractory	100	100	100	100	85	100	0
BM P051910(I)	refractory	100	0	N/A	N/A	80	85	0
PB 74165E59	refractory	100	100	100	100	98	100	0
BM 74165E59	refractory	100	100	100	100	94	100	0
BM CML007	refractory	0	86	0	0	0	0	0

Primary CML samples were stained with anti-EphA3 antibody, PE-conjugated anti-CD34, PEcy5-conjugated anti-CD38 and APC-conjugated anti-CD123 for flow cytometry analysis. For ADCC assays, 10 μg/mL KB004 was added to primary patient sample target cells together with PBMC effector cells from healthy volunteers at an effector-to-target cell ratio of 100:1 for 16 hours. For apoptosis assays, KB004 at 10 μg/mL or Imatinib at 10 μg/mL (17 μM) was added to target cells for 24 hours. Data for 9 $EphA3^+$ CML samples and 2 representative $EphA3^-$ samples are shown. N/A = Not applicable. N/D = Not determined. Samples are from bone marrow aspirates (BM) or peripheral blood (PB).

CONCLUSIONS

- EphA3 is expressed on the cell surface in a significant number of samples from patients with AML, CML, MDS, MPN and MM.
- Cells with leukemic stem cell phenotype ($CD34^+$ $CD38^-$ $CD123^+$) express EphA3.
- KB004 inhibits colony formation from long-term culture-initiating cells (LTC-IC) from AML stem cells and does not inhibit normal human bone marrow LTC-IC.
- KB004 inhibits AML progenitor cell colony formation and does not inhibit normal human bone marrow progenitor cell colony formation.
- KB004 has potent cell-killing activity against EphA3-positive hematologic malignancies.
 - Direct induction of apoptosis
 - Enhanced ADCC activity
- KB004 kills EphA3 positive cells from Imatinib-refractory CML patients by apoptosis and ADCC.
- Opportunity to target leukemic stem cells and blasts with a potent and selective antibody while sparing normal bone marrow cells.
- A Phase 1 first-in-man study will be conducted at MD Anderson Cancer Center and Moffitt Cancer Center.

REFERENCES

Boyd *et al.* (1992) J. Biol. Chem. **267**:3262-3267.
Wicks *et al.* (1992) Proc. Natl. Acad. Sci. U S A. **89**:1611-1615.
Smith *et al.* (1994) J. Biol. Chem. **279**: 9522-9531.
Mori *et al.* (2007) Cytotechnology. **55**:109-14.