Lovastatin Improves Stool Form in *Methanobrevibacter smithii* Colonized Rats with Constipation

Walter Morales[1], Emily Marsh[1], Allen Yu[1], Zachary Marsh[1], Stacy Weitsman[1], Gillian M Barlow[1], Ali Rezaie[1], Christopher Chang[1], Vince Wacher[2], Mark Pimentel[1]

1. GI Motility Program, Cedars-Sinai Medical Center, Los Angeles, CA, United States
2. Synthetic Biologics, Inc., Rockville, Maryland

BACKGROUND

Evidence suggests that methane and colonization with *Methanobrevibacter smithii* (*M. smithii*) may be important in the pathogenesis of constipation and constipation-predominant irritable bowel syndrome (C-IBS). It has been demonstrated that direct *in vivo* infusion of methane into dogs slows intestinal transit by 60%.

Lovastatin is a HMG-CoA reductase inhibitor produced by fungal species of the genus *Aspergillus*. HMG-CoA reductase inhibitors act on the rate-limiting enzyme in the mevalonate pathway important in cholesterol production in humans. In addition to this effect, lovastatin has specifically been shown to reduce the production of methane by inhibition of methanogenesis in *M. smithii*.

AIM

In this study, we investigate the effects of lovastatin in a rat model of diet-induced *M. smithii* proliferation and resulting constipation.

METHODS

30 adult, male Sprague-Dawley rats were placed on a high-fat diet (60.3% kcal from fat, Teklad high-fat diet TD.06414, Harlan Laboratories Inc, Madison, WI) for 7 weeks. The rats were assessed for increased *M. smithii* by qPCR before and after the diet, and then divided into 3 groups. Group 1 was given lovastatin in its lactone form, Group 2 was given lovastatin hydroxy acid (each 1.5 mg/rat), and Group 3 was gavaged with a placebo. Each group was gavaged daily for 10 days. Three day stool collections were performed to assess average stool wet weight and daily variability prior to commencing the high-fat diet, after 7 weeks of high-fat diet, and the final days of the lovastatin gavage (still on high-fat diet). On day 10 of the gavage, rats were euthanized and DNA was extracted from contents of ligated bowel segments (duodenum, jejunum, ileum, cecum and left colon). qPCR was performed using primers for total luminal bacteria and *M. smithii*.

RESULTS

Confirming previous studies, high-fat diet augmented stool *M. smithii* colonization in Sprague-Dawley rats ($7.58 \times 10^4 \pm 6.62 \times 10^4$ cfu/mL at baseline to $2.60 \times 10^5 \pm 1.95 \times 10^5$ after 7 weeks of high-fat) (P<0.01) (Figure 1). This was coupled with a reduction in the stool wet-weights (62.4% at baseline to 48.6% after 7 weeks) (P<0.01) (Figure 2). At this point rats were divided into 3 groups. With respect to the total bacteria by qPCR, levels were not different between placebo and either lovastatin group. For *M. smithii*, the ratio of *M. smithii* to total bacteria was reduced in the ileum of rats given the lovastatin lactone but not hydroxy acid. *M. smithii* levels in the colon were unaffected (Figure 3). Most importantly, there was an increase in stool wet weight noted in rats receiving lovastatin lactone gavage (Figure 4).

Study Design:

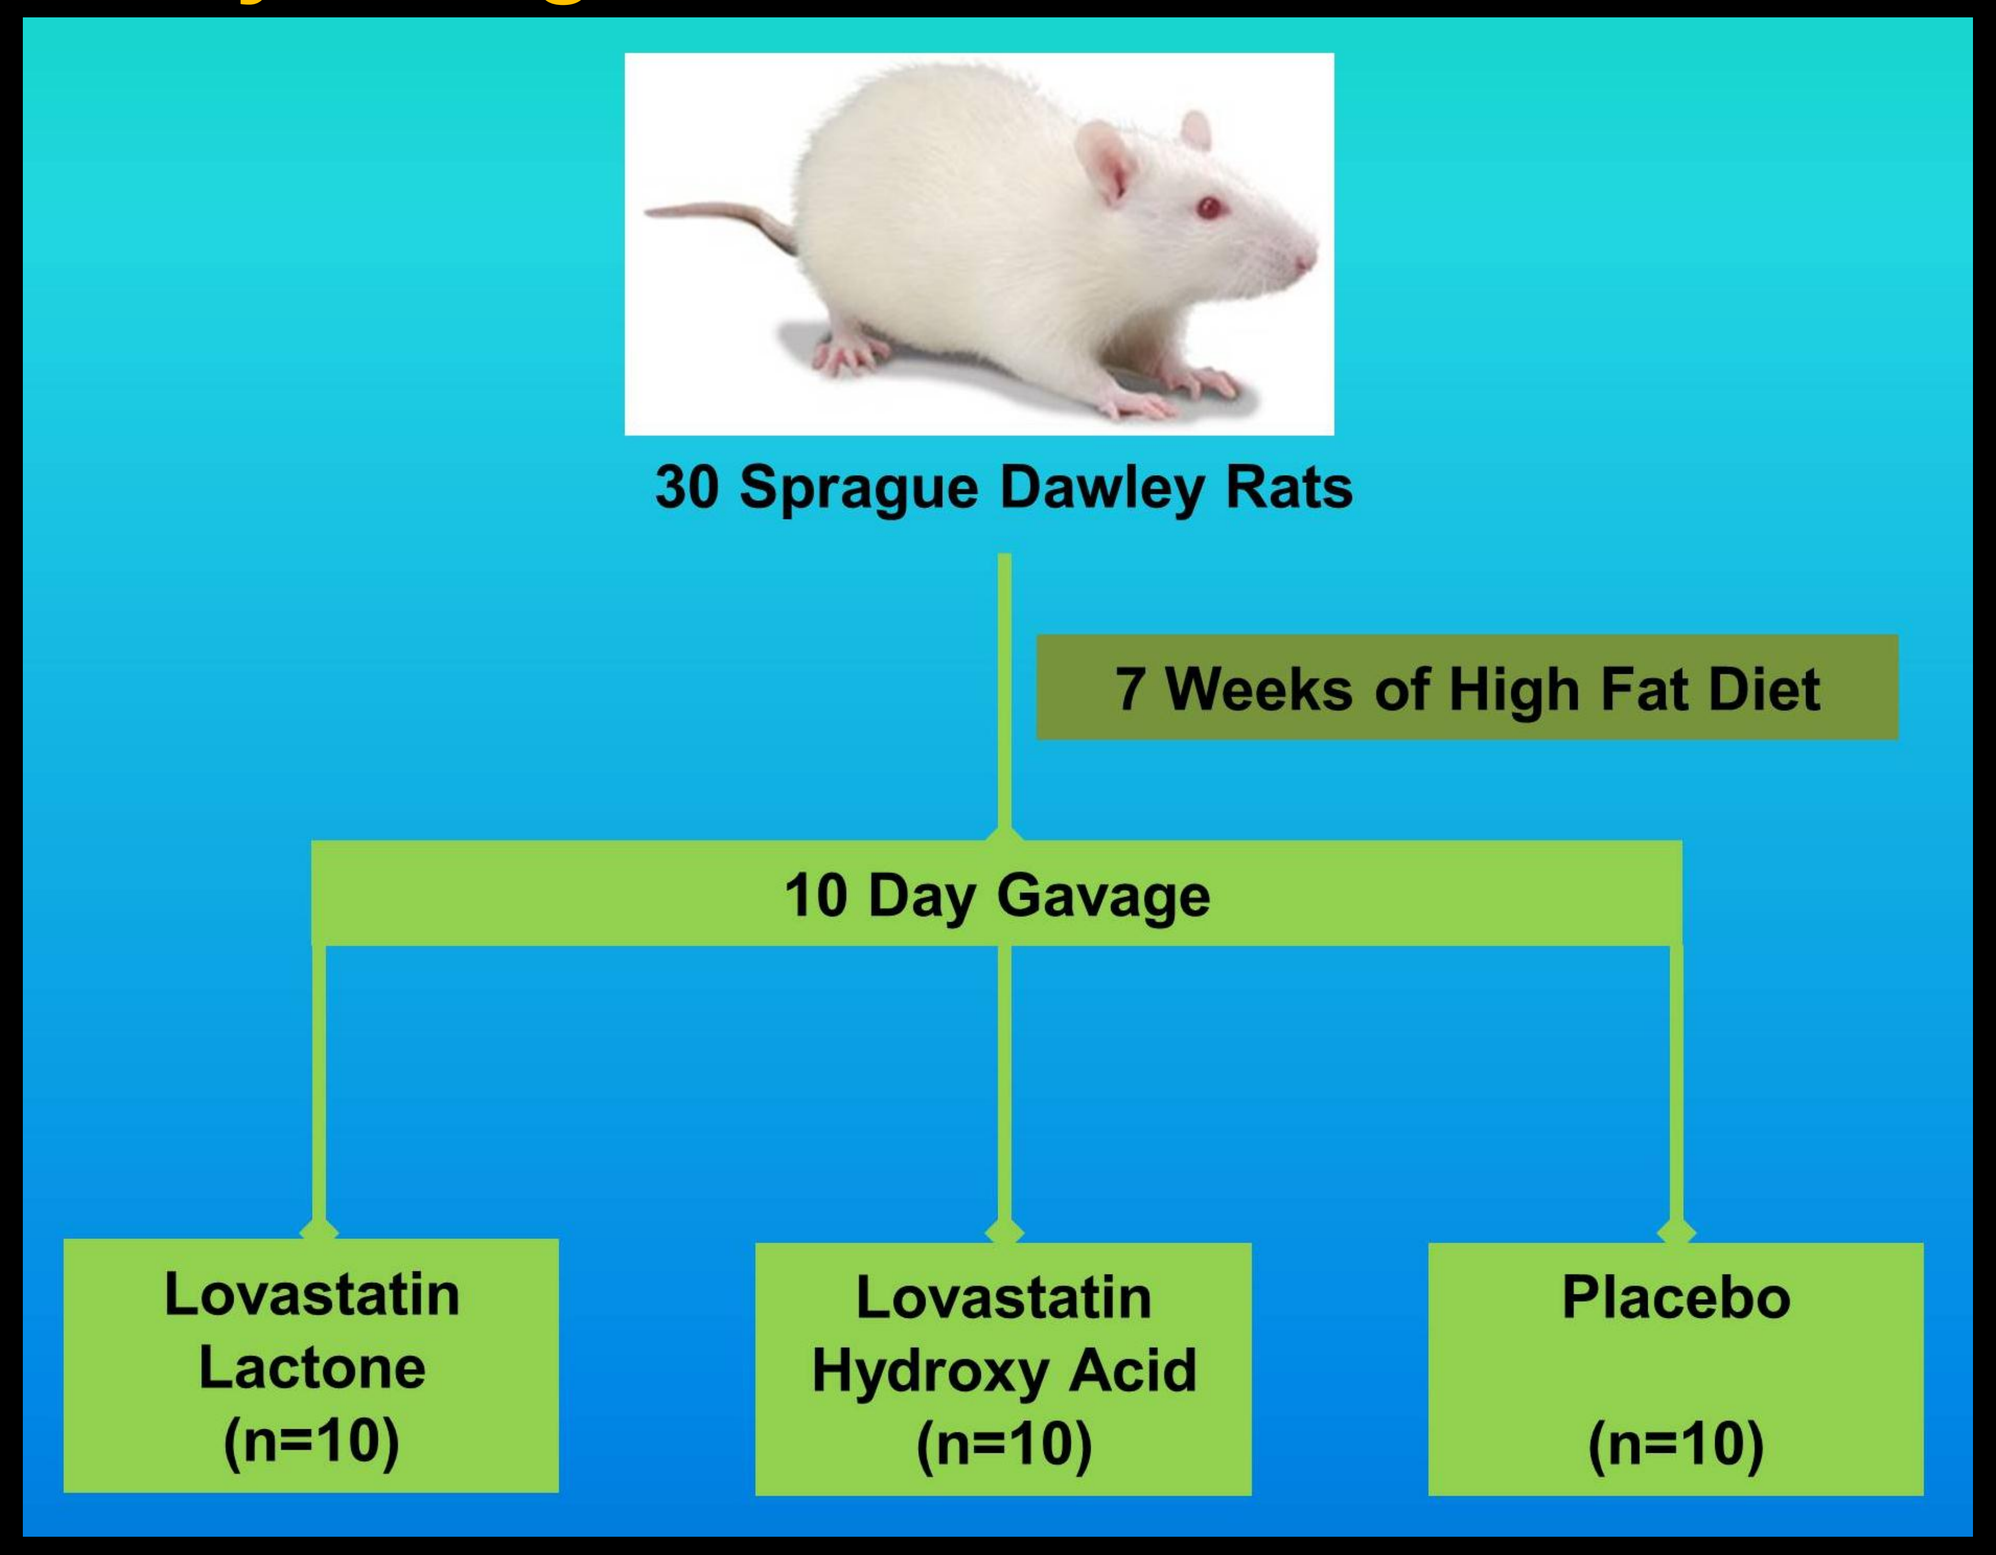
30 Sprague Dawley Rats
7 Weeks of High Fat Diet
10 Day Gavage
Lovastatin Lactone (n=10)
Lovastatin Hydroxy Acid (n=10)
Placebo (n=10)

Figure 1: Augmented stool *M. smithii* after 7 weeks on a high-fat diet (HF)

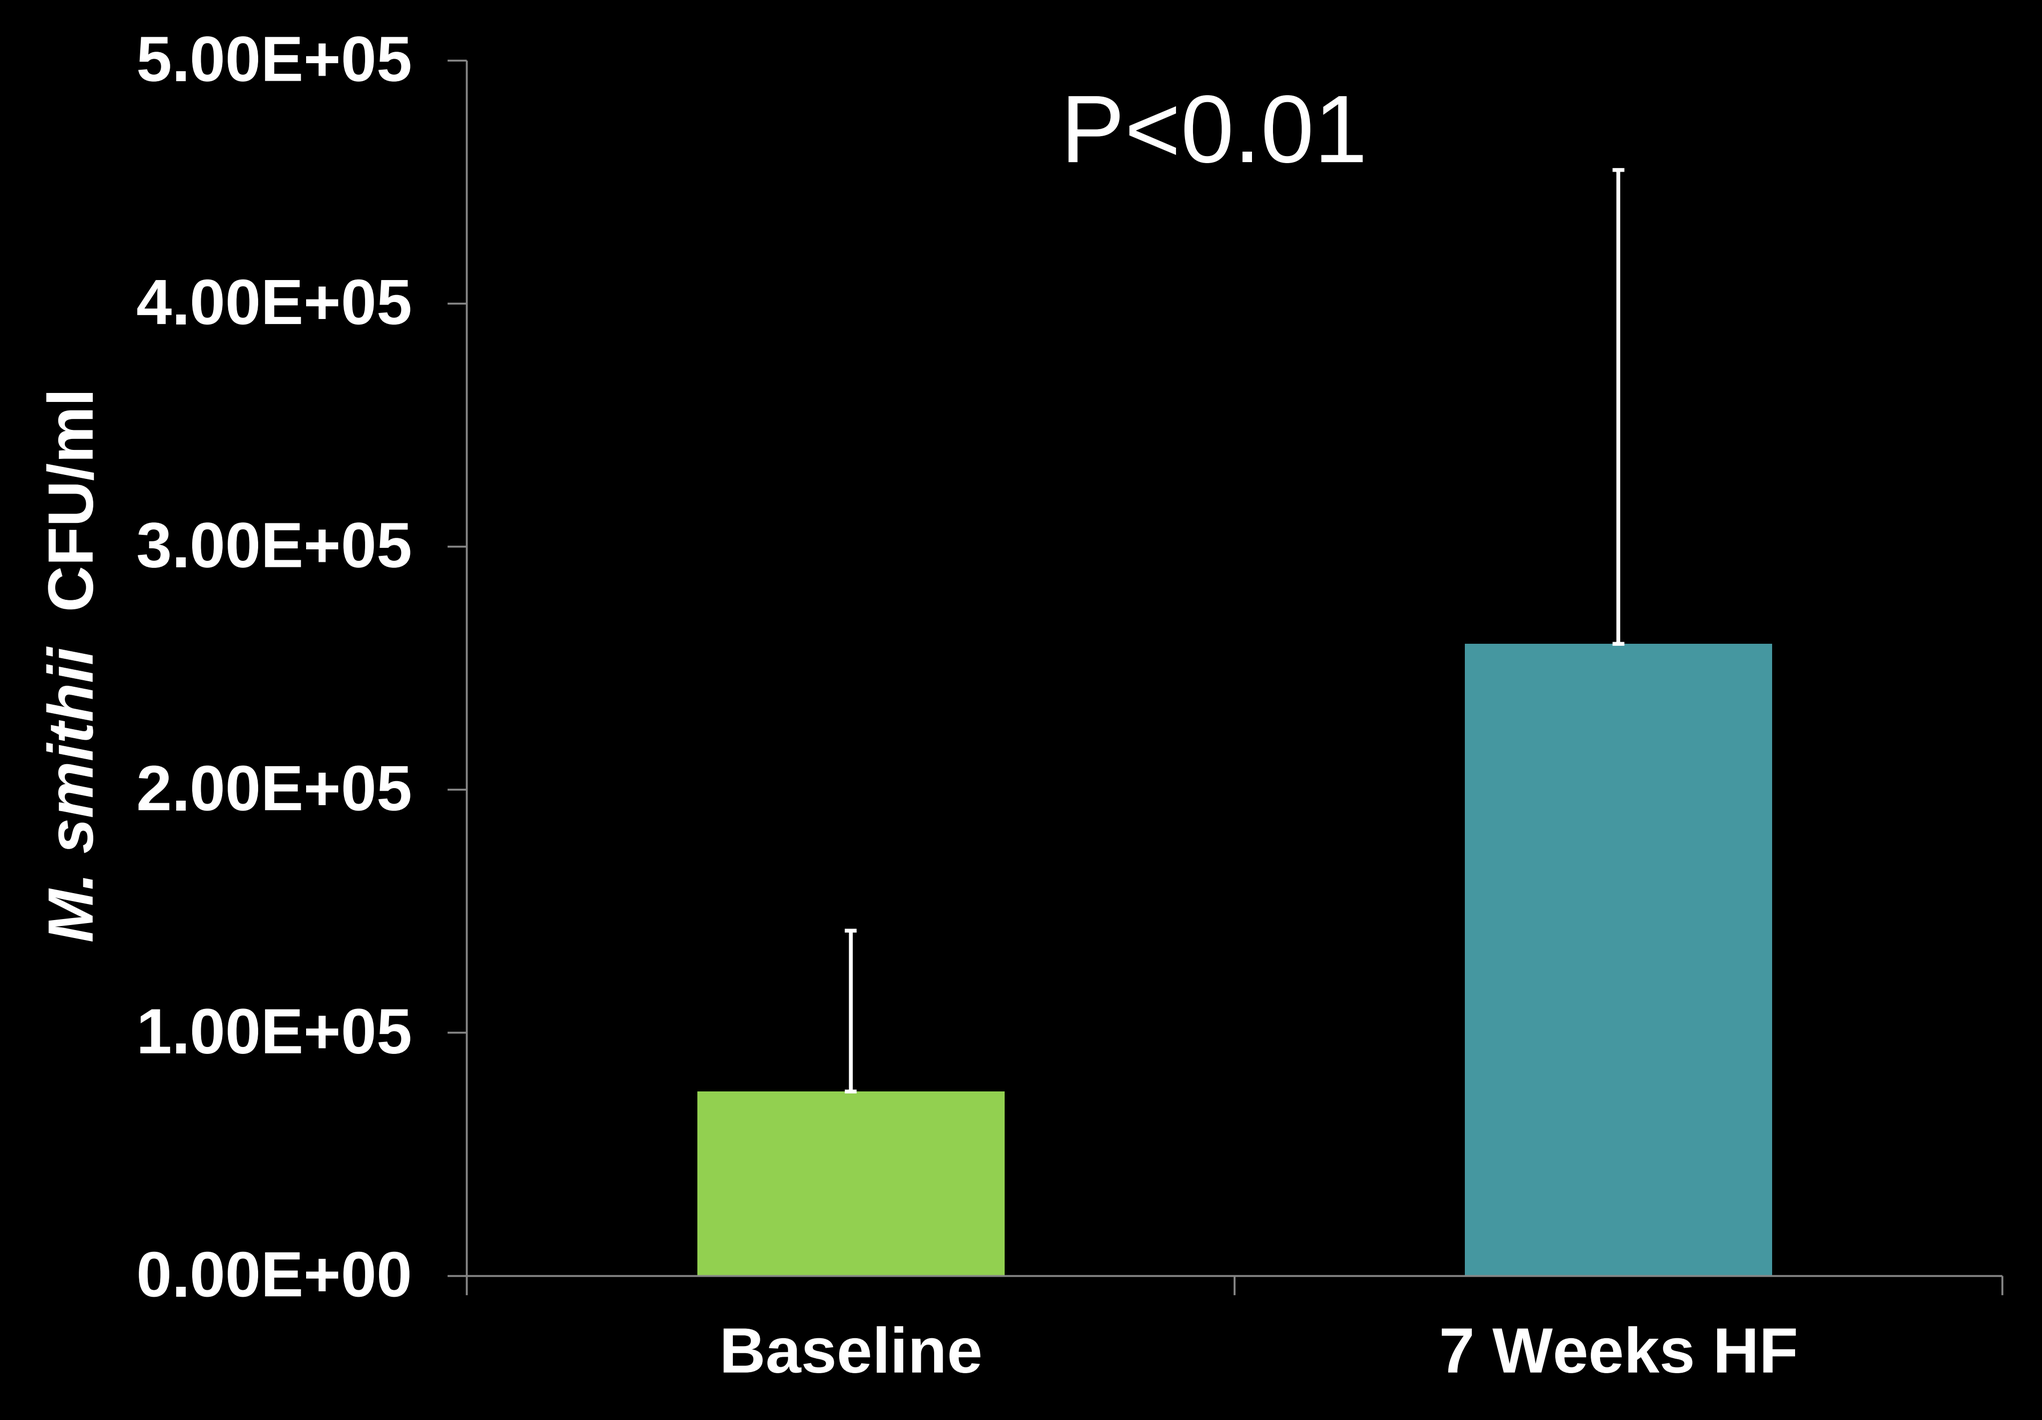
P<0.01
M. smithii CFU/ml
5.00E+05
4.00E+05
3.00E+05
2.00E+05
1.00E+05
0.00E+00
Baseline
7 Weeks HF

Figure 2: Reduction in stool wet-weights after 7 weeks on a high-fat diet (HF)

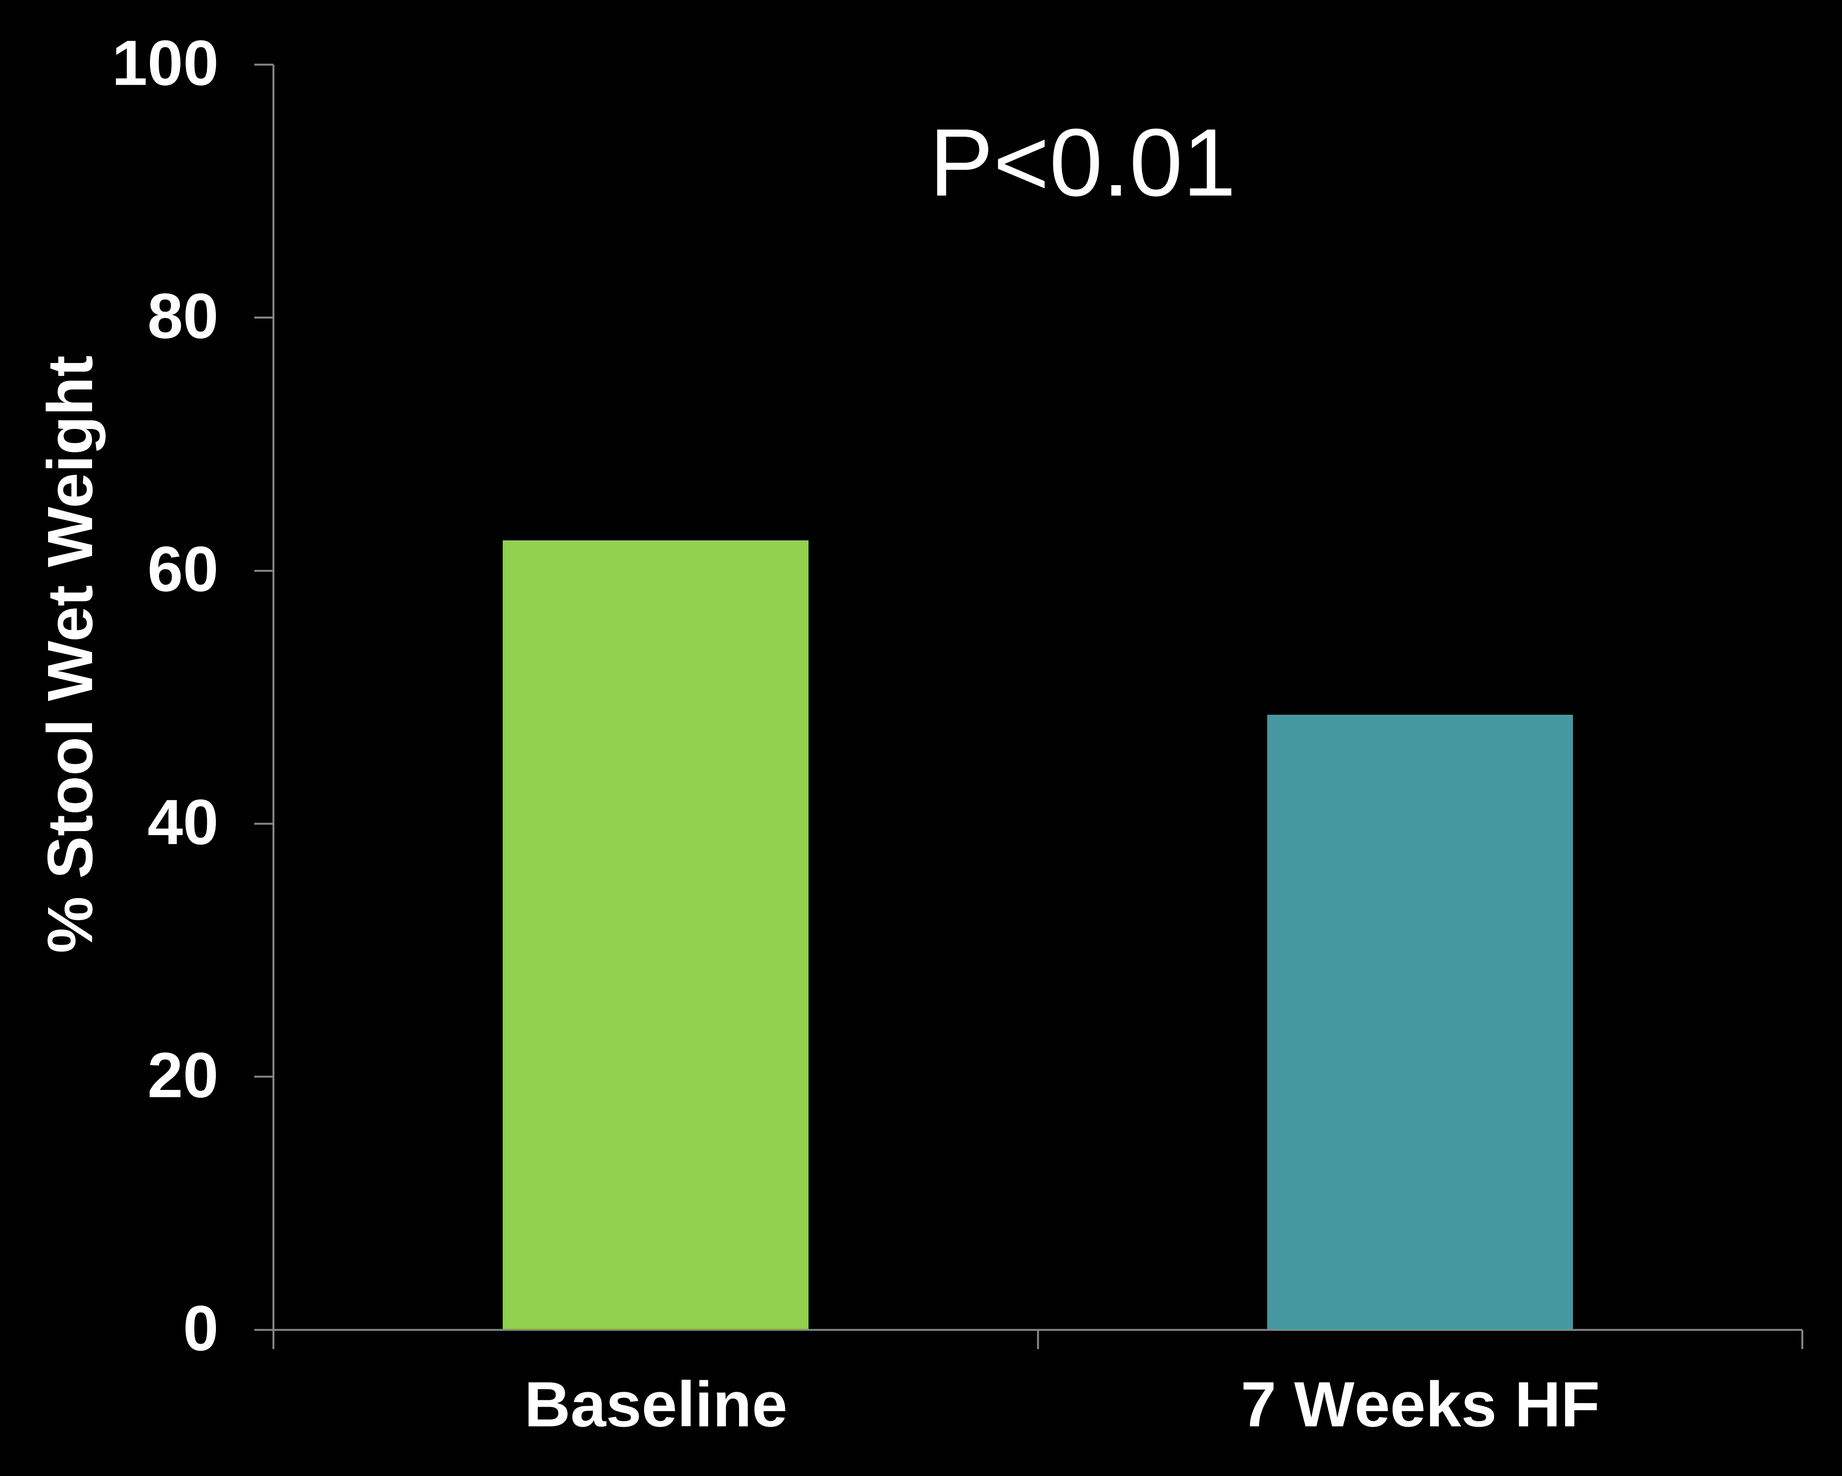
P<0.01
% Stool Wet Weight
100
80
60
40
20
0
Baseline
7 Weeks HF

Figure 3: After lovastatin gavage: (a) Ileal ratio of *M. smithii* to total bacteria via qPCR (b) Colonic ratio of *M. smithii* to total bacteria via qPCR

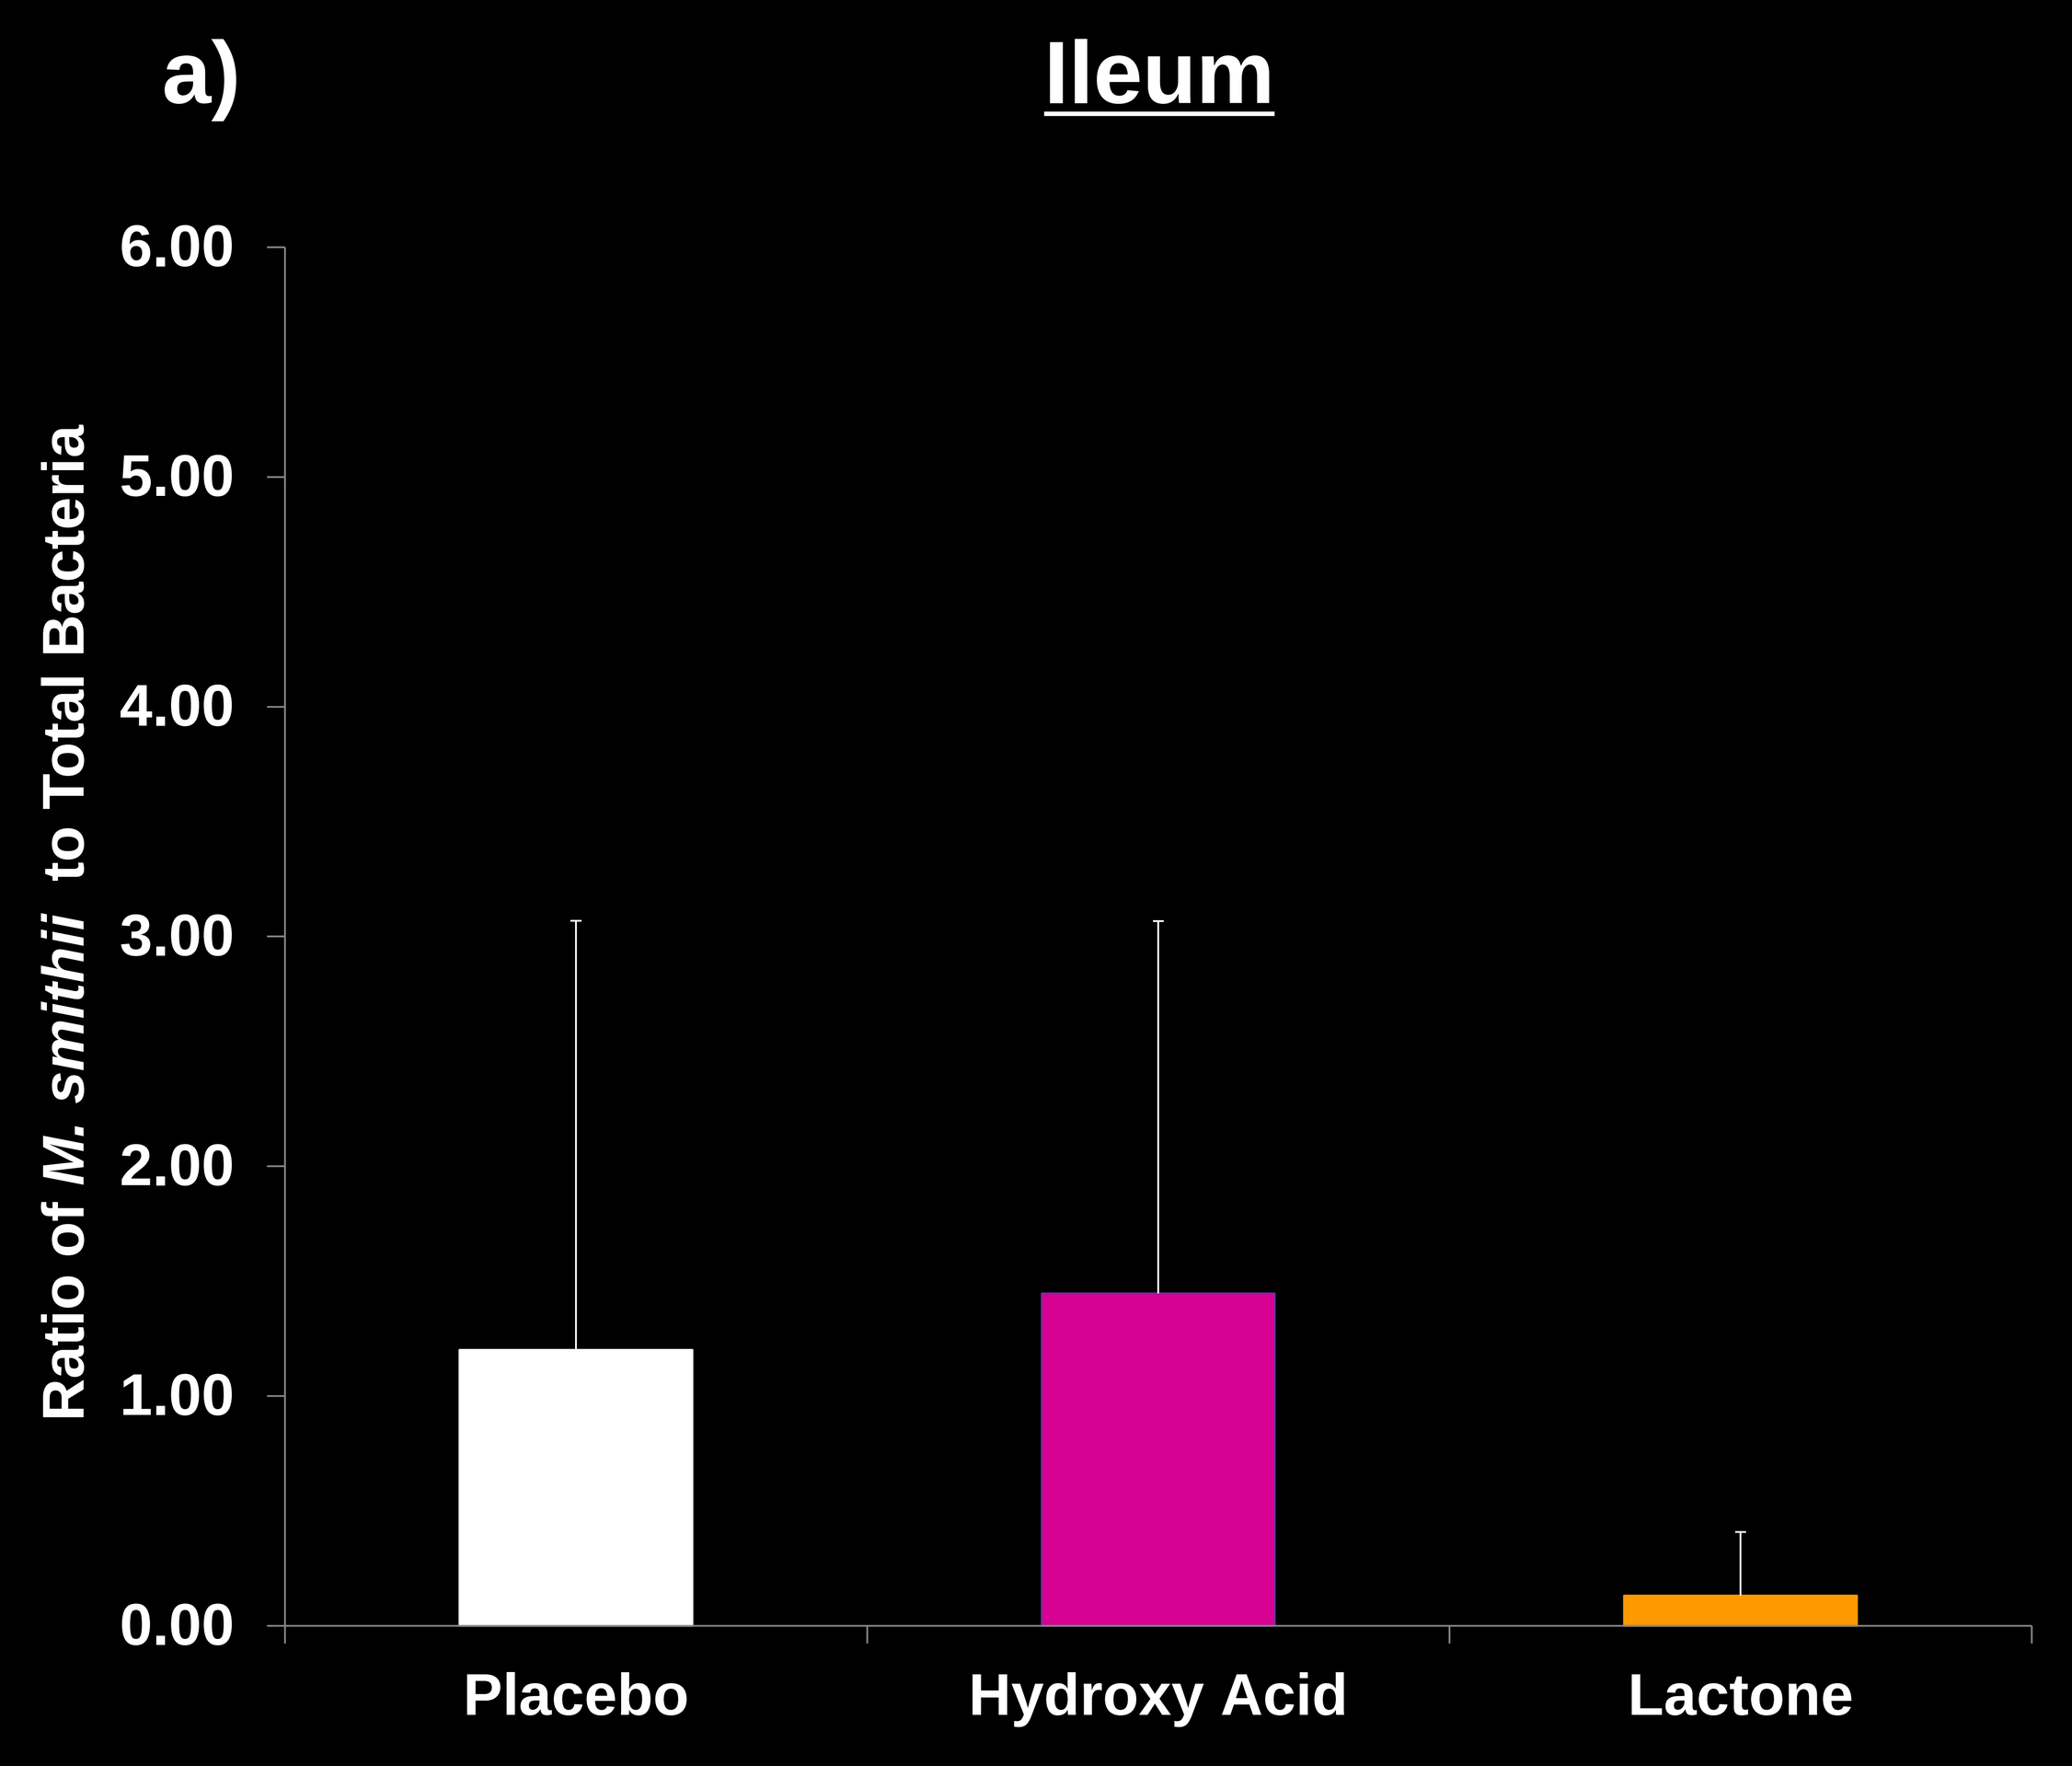
a)
Ileum
Ratio of M. smithii to Total Bacteria
6.00
5.00
4.00
3.00
2.00
1.00
0.00
Placebo
Hydroxy Acid
Lactone

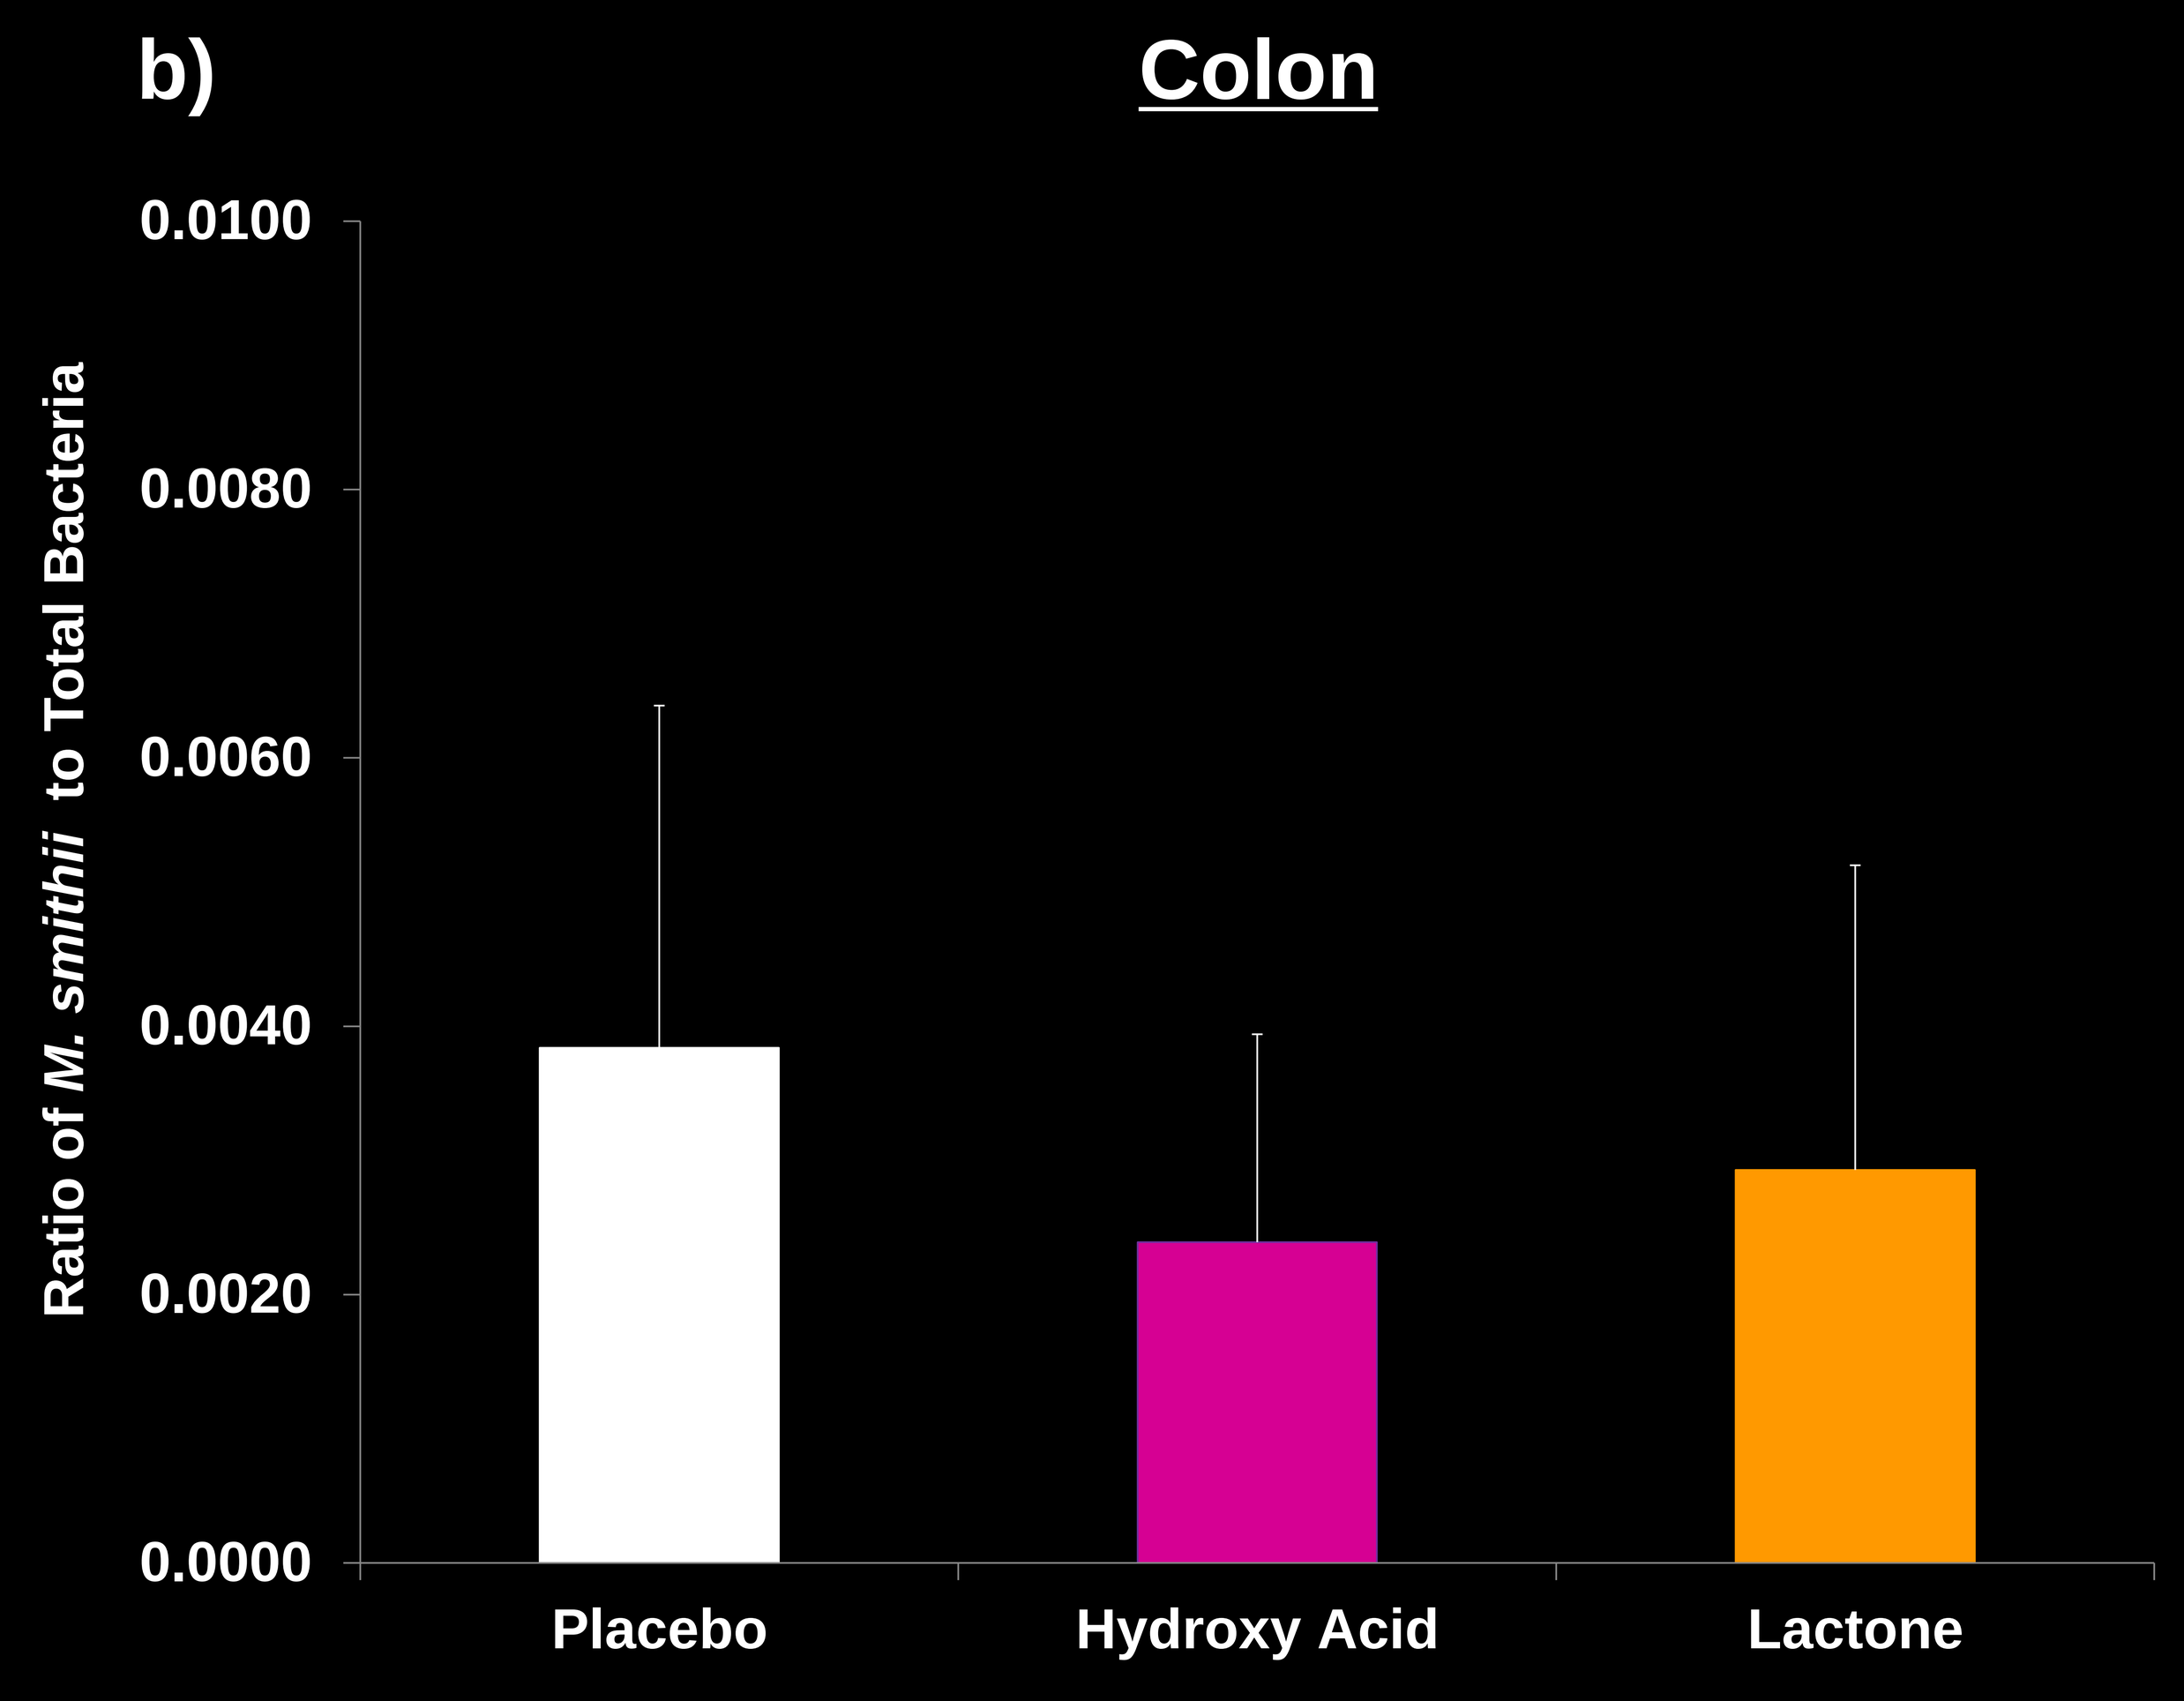
b)
Colon
Ratio of M. smithii to Total Bacteria
0.0100
0.0080
0.0060
0.0040
0.0020
0.0000
Placebo
Hydroxy Acid
Lactone

Figure 4: Changes in stool wet weight during high-fat diet (HF) and after 10 day gavage

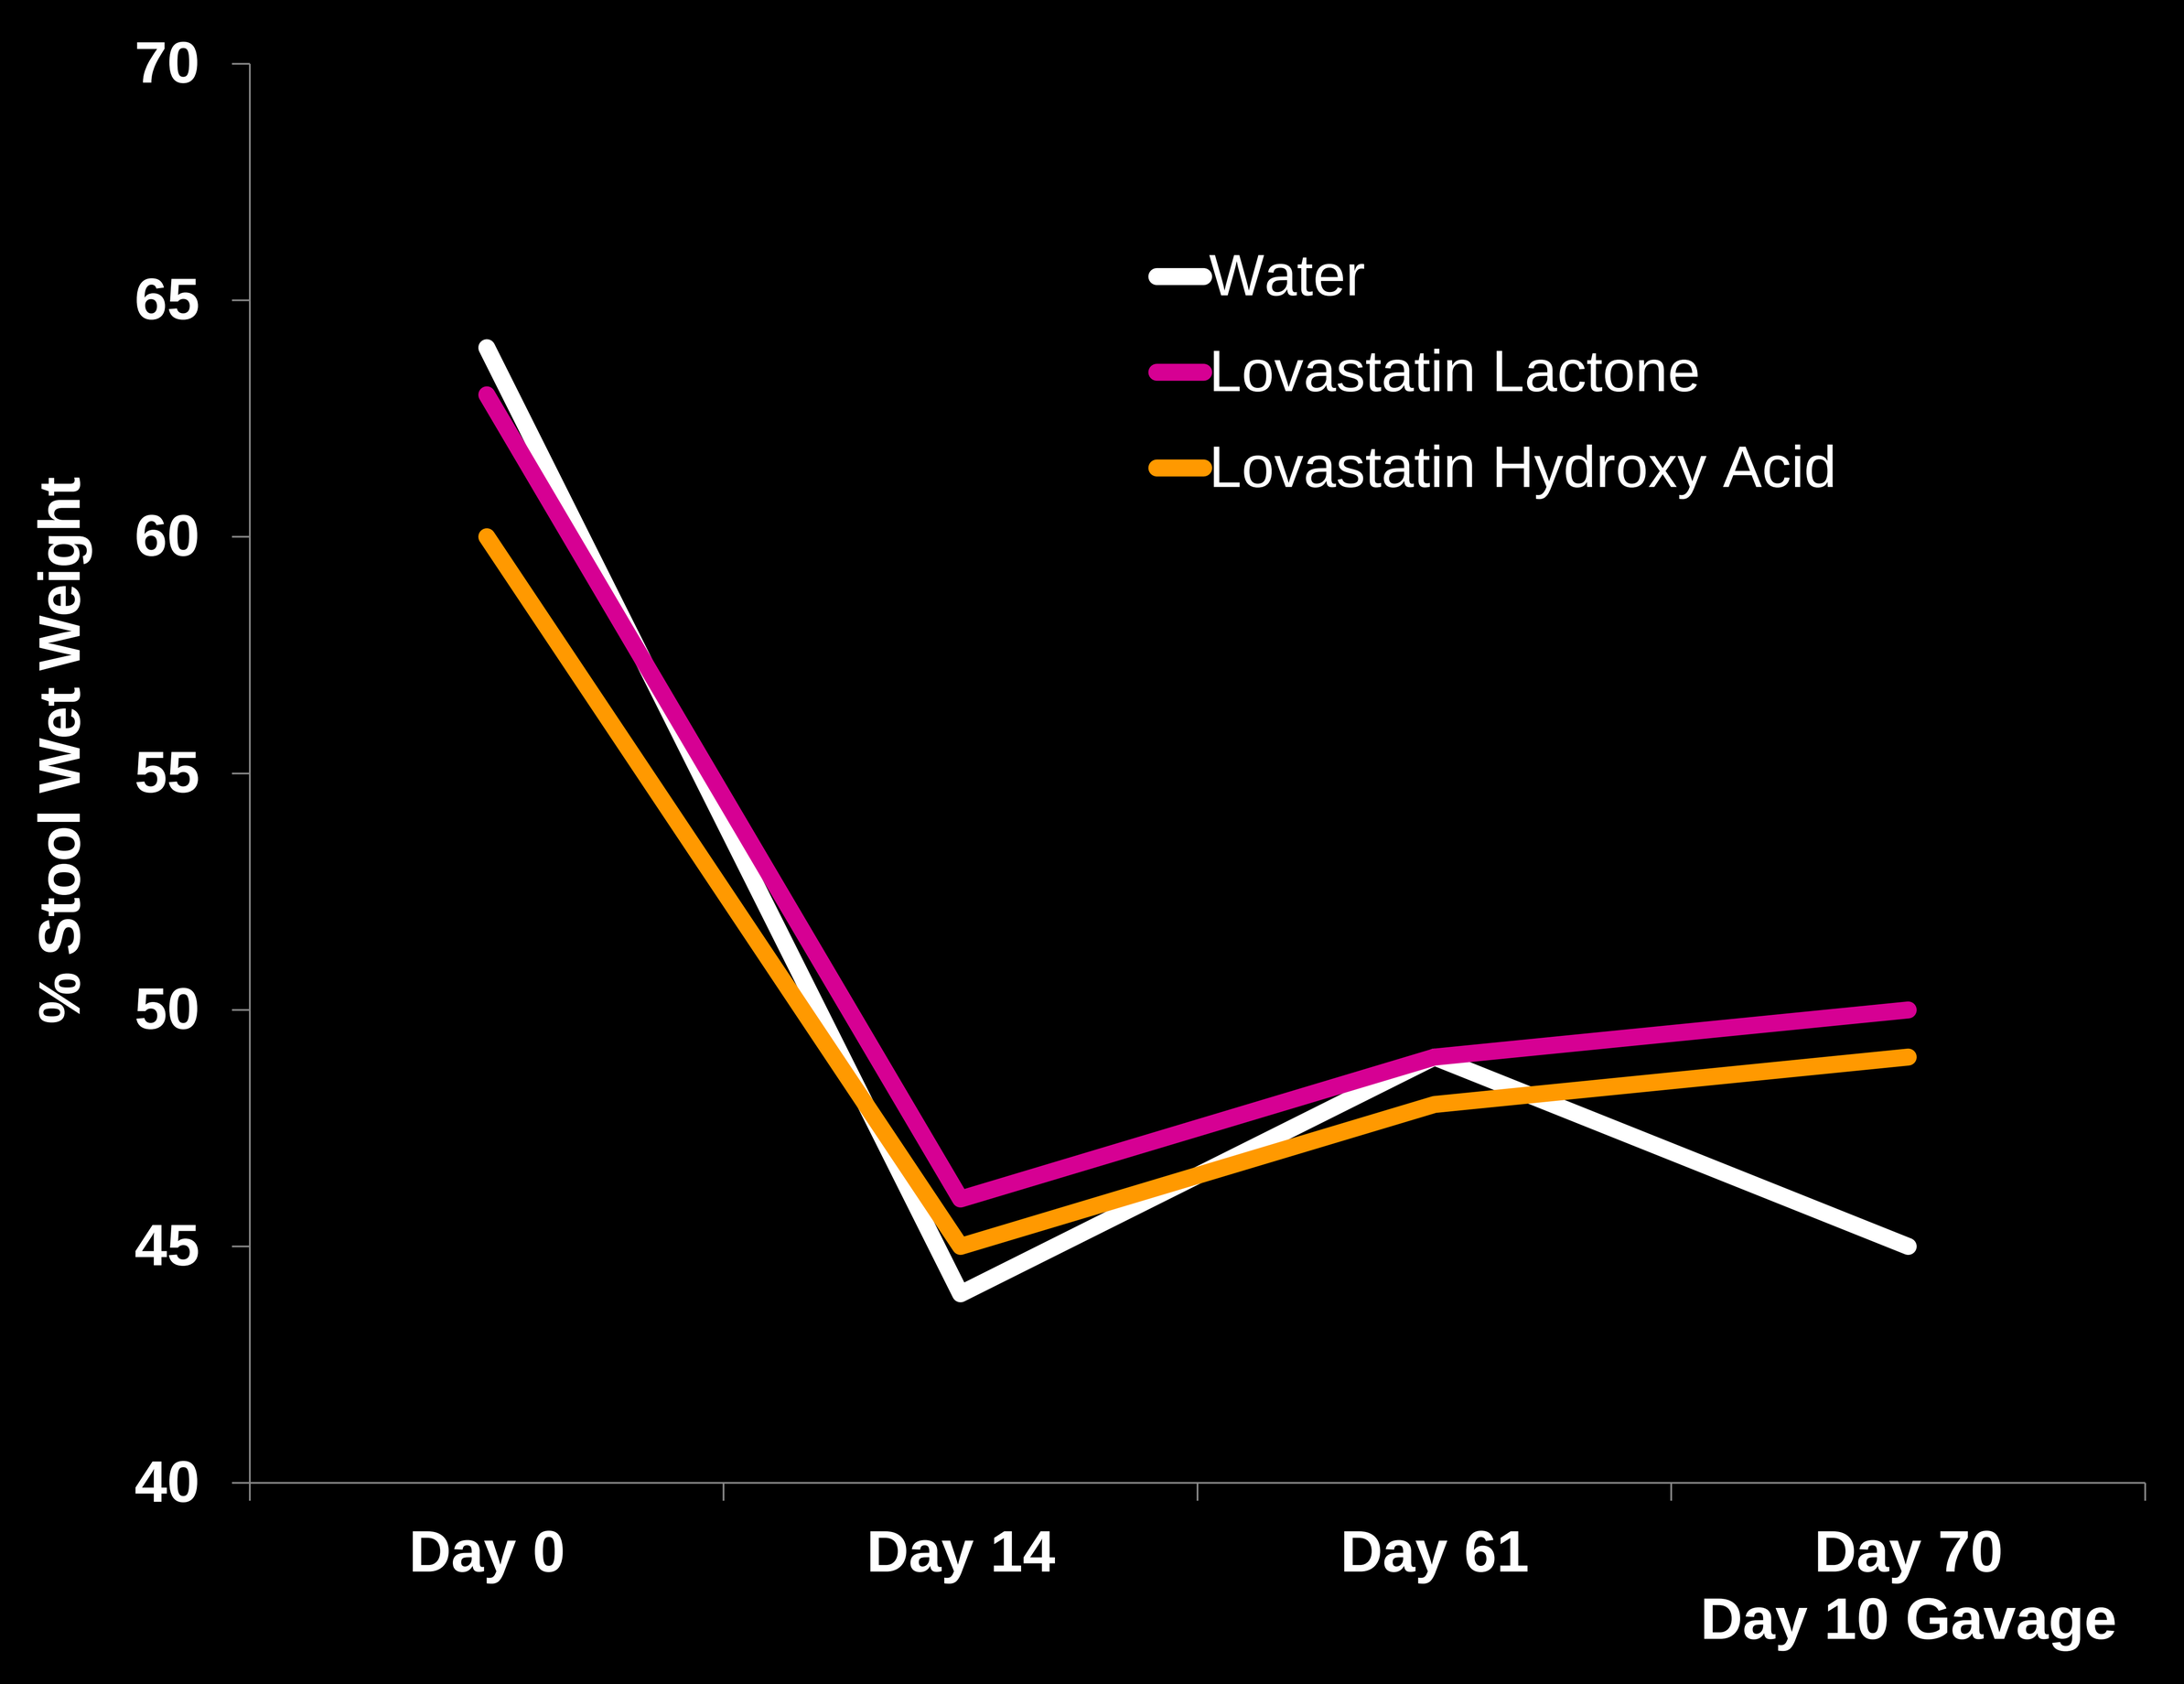
% Stool Wet Weight
70
65
60
55
50
45
40
Water
Lovastatin Lactone
Lovastatin Hydroxy Acid
Day 0
Day 14
Day 61
Day 70
Day 10 Gavage

CONCLUSIONS

In this study, we show that:

1. Lovastatin improved stool wet weight in rats with augmented *M. smithii*.
2. Lovastatin lactone reduced *M. smithii* levels only in the ileum.
3. It is possible that the use of lovastatin could improve constipation by alterations in *M. smithii*.

This research was sponsored by Synthetic Biologics, Inc.