Gut Antibiotic Inactivation by Oral Beta-Lactamase Therapy Protects the Microbiome from IV and Oral Antibiotics and Reduces Propagation of Antibiotic Resistance in Large Animal Dysbiosis Models

Sheila Connelly[1], Christian Furlan-Freguia[1], Brian Fanelli[2], Nur A. Hasan[2], Rita R. Colwell[2], Michael Kaleko[1]
[1]Synthetic Biologics, Inc., Rockville, MD, USA, [2]CosmosID, Inc., Rockville, MD, USA

sconnelly@syntheticbiologics.com

ABSTRACT

Background: Exposure of the gut microbiota to antibiotics can damage the microbiome and lead to the propagation of antibiotic resistance. SYN-004 (ribaxamase) is a clinical stage oral beta-lactamase for use with parenteral beta-lactam antibiotics, including penicillins and cephalosporins, intended to preserve the microbiome by degrading antibiotics in the intestine. A proof-of-concept study assessing ribaxamase efficacy in patients receiving IV ceftriaxone met its primary endpoint of significantly reducing *C. difficile* infection. Ribaxamase protected the gut microbiome from IV ceftriaxone and reduced antibiotic resistance in a porcine model of antibiotic-mediated dysbiosis. The ability of delayed release ribaxamase formulations to protect the canine gut microbiome from oral amoxicillin was evaluated.

Methods: Delayed-release ribaxamase formulations, named SYN-007, were engineered for dissolution in the lower small intestine distal to the site of oral antibiotic absorption. Multiple SYN-007 formulations were evaluated in vitro and three chosen for in vivo testing. Dogs received amoxicillin (80 mg/kg, PO, TID) +/- SYN-007 (10 mg, PO, TID) for 5 days. Serum amoxicillin levels were measured after the first and last antibiotic doses by LC/MS/MS. Fecal DNA whole genome shotgun sequence analyses were performed with CosmosID metagenomics software.

Results: With one SYN-007 formulation, serum amoxicillin levels were not significantly different from amoxicillin controls for both the first and last doses indicating that SYN-007 did not degrade the antibiotic prior to its systemic absorption. Exposure of the gut microbiota to amoxicillin alone resulted in loss of some bacterial species and overgrowth of others, and emergence and propagation of antibiotic resistance genes. The genes included encoded beta-lactamases, conferring resistance to amoxicillin, components of multi-drug efflux pumps, and genes providing resistance to a broad range of antibiotic classes. In the presence of amoxicillin+SYN-007, both microbiome disruption and antibiotic resistance were reduced.

Conclusions: SYN-007 diminished amoxicillin-mediated microbiome disruption and mitigated emergence and propagation of antibiotic resistance genes. Antibiotic inactivation represents a new treatment paradigm for preservation of the gut microbiome to reduce opportunistic infections and antibiotic resistance. SYN-007 has the potential to expand microbiome protection to include oral as well as IV beta-lactam antibiotics.

BACKGROUND

IV beta-lactam antibiotics, including cephalosporins, are excreted via the bile into the intestine where they can disrupt the intestinal microbiota and potentially lead to outgrowth of pathogens like *Clostridium difficile*. SYN-004 (ribaxamase) is a clinical stage, oral beta-lactamase enzyme therapy for use with IV beta-lactam antibiotics designed to preserve the gut microbiome by degrading residual antibiotics in the GI tract. A phase 2b study met its primary endpoint of significantly reducing *C. difficile* infection (CDI) in patients treated with ceftriaxone and ribaxamase.

The ribaxamase clinical formulation consists of enteric-coated enzyme pellets engineered to protect the enzyme from stomach acid with enzyme release occuring at pH >5.5 in the upper small intestine. As expected, use of the clinical formulation with oral amoxicillin in pigs prevented systemic absorption of the antibotic, presumably due to degradation of the antibiotic prior to absorption.

Novel formulations of ribaxamase, named SYN-007, intended to release enzyme in the GI tract at a site distal to oral antibiotic absorption, were developed and evaluated *in vitro* and in dogs. One SYN-007 formulation did not interfere with amoxicillin absorption and protected the gut microbiome from antibiotic collateral damage, including maintaining microbiota diversity and reducing propagation of antibiotic resistance in dogs.

RESULTS

Distal Release Ribaxamase Formulations

Distal-release formulations of the beta-lactamase, ribaxamase, were engineered for dissolution in the lower small intestine distal to the site of oral antibiotic absorption.

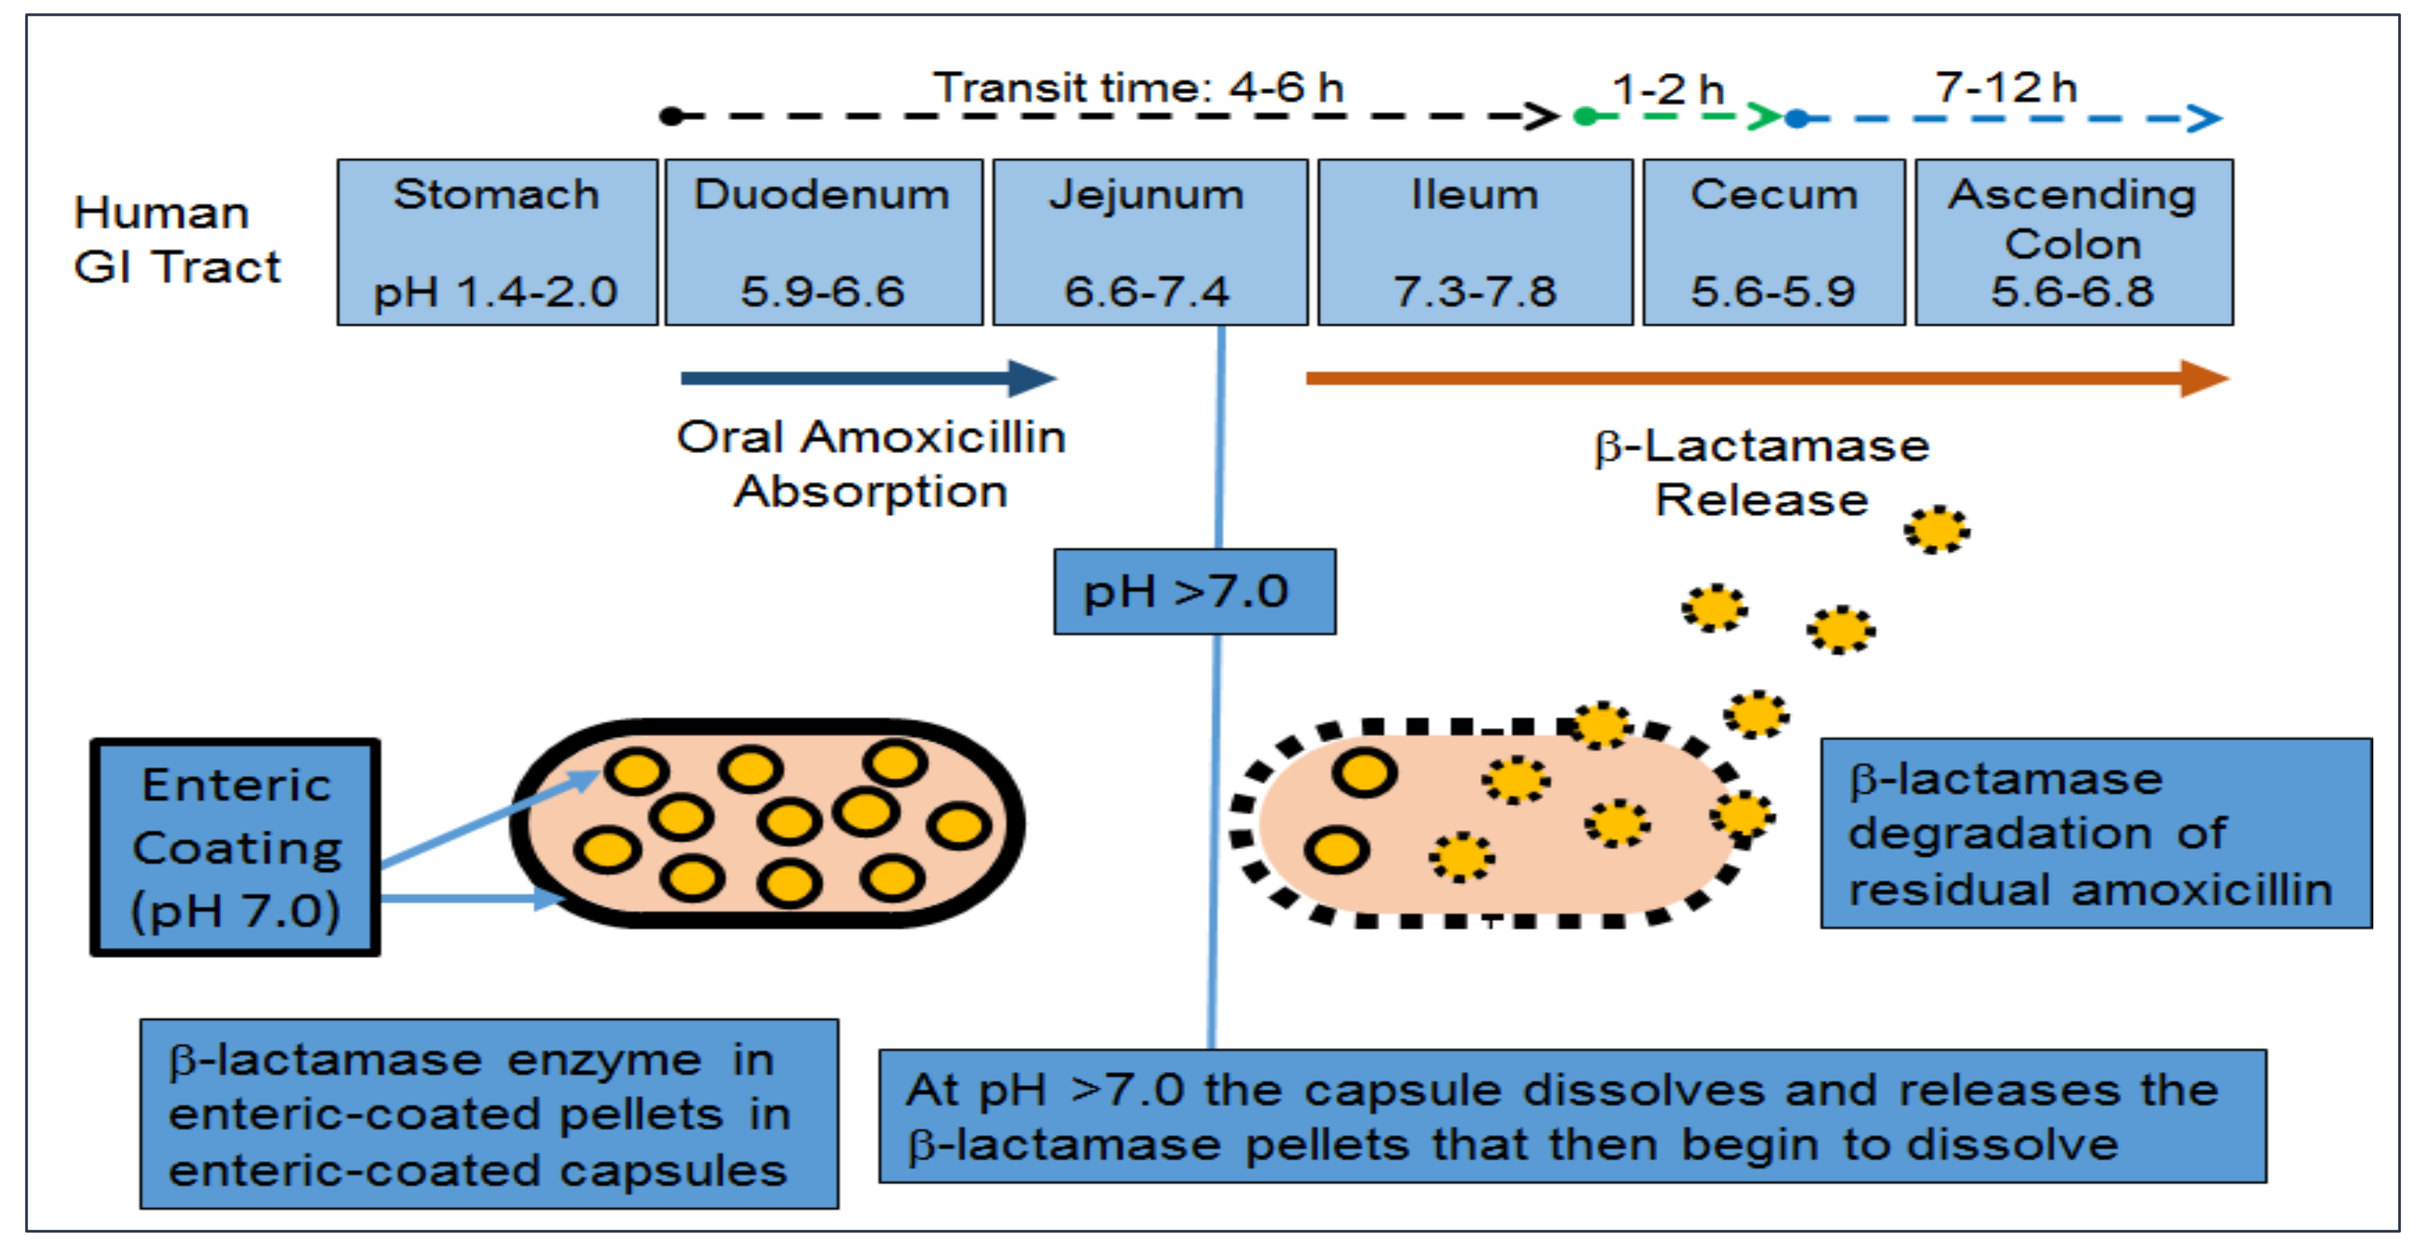
Transit time: 4-6 h
1-2 h
7-12 h
Human GI Tract
Stomach pH 1.4-2.0
Duodenum 5.9-6.6
Jejunum 6.6-7.4
Ileum 7.3-7.8
Cecum 5.6-5.9
Ascending Colon 5.6-6.8
Oral Amoxicillin Absorption
β-Lactamase Release
pH >7.0
Enteric Coating (pH 7.0)
β-lactamase degradation of residual amoxicillin
β-lactamase enzyme in enteric-coated pellets in enteric-coated capsules
At pH >7.0 the capsule dissolves and releases the β-lactamase pellets that then begin to dissolve

Three distal-release formulations were produced. Formulations 1 and 3 were composed of enteric-coated enzyme pellets within enteric-coated capsules. Formulation 2 contained enteric-coated pellets in uncoated capsules. Dissolution was tested *in vitro.* Capsules were held in 0.1N HCl (pH 1.1) for 2 hrs to simulate conditions in the stomach, 2 hrs at pH 5.5, conditions of the upper small intestine, and 4 hrs at pH 7.1, conditions of the lower small intestine/colon.

Distal-Release Formulations

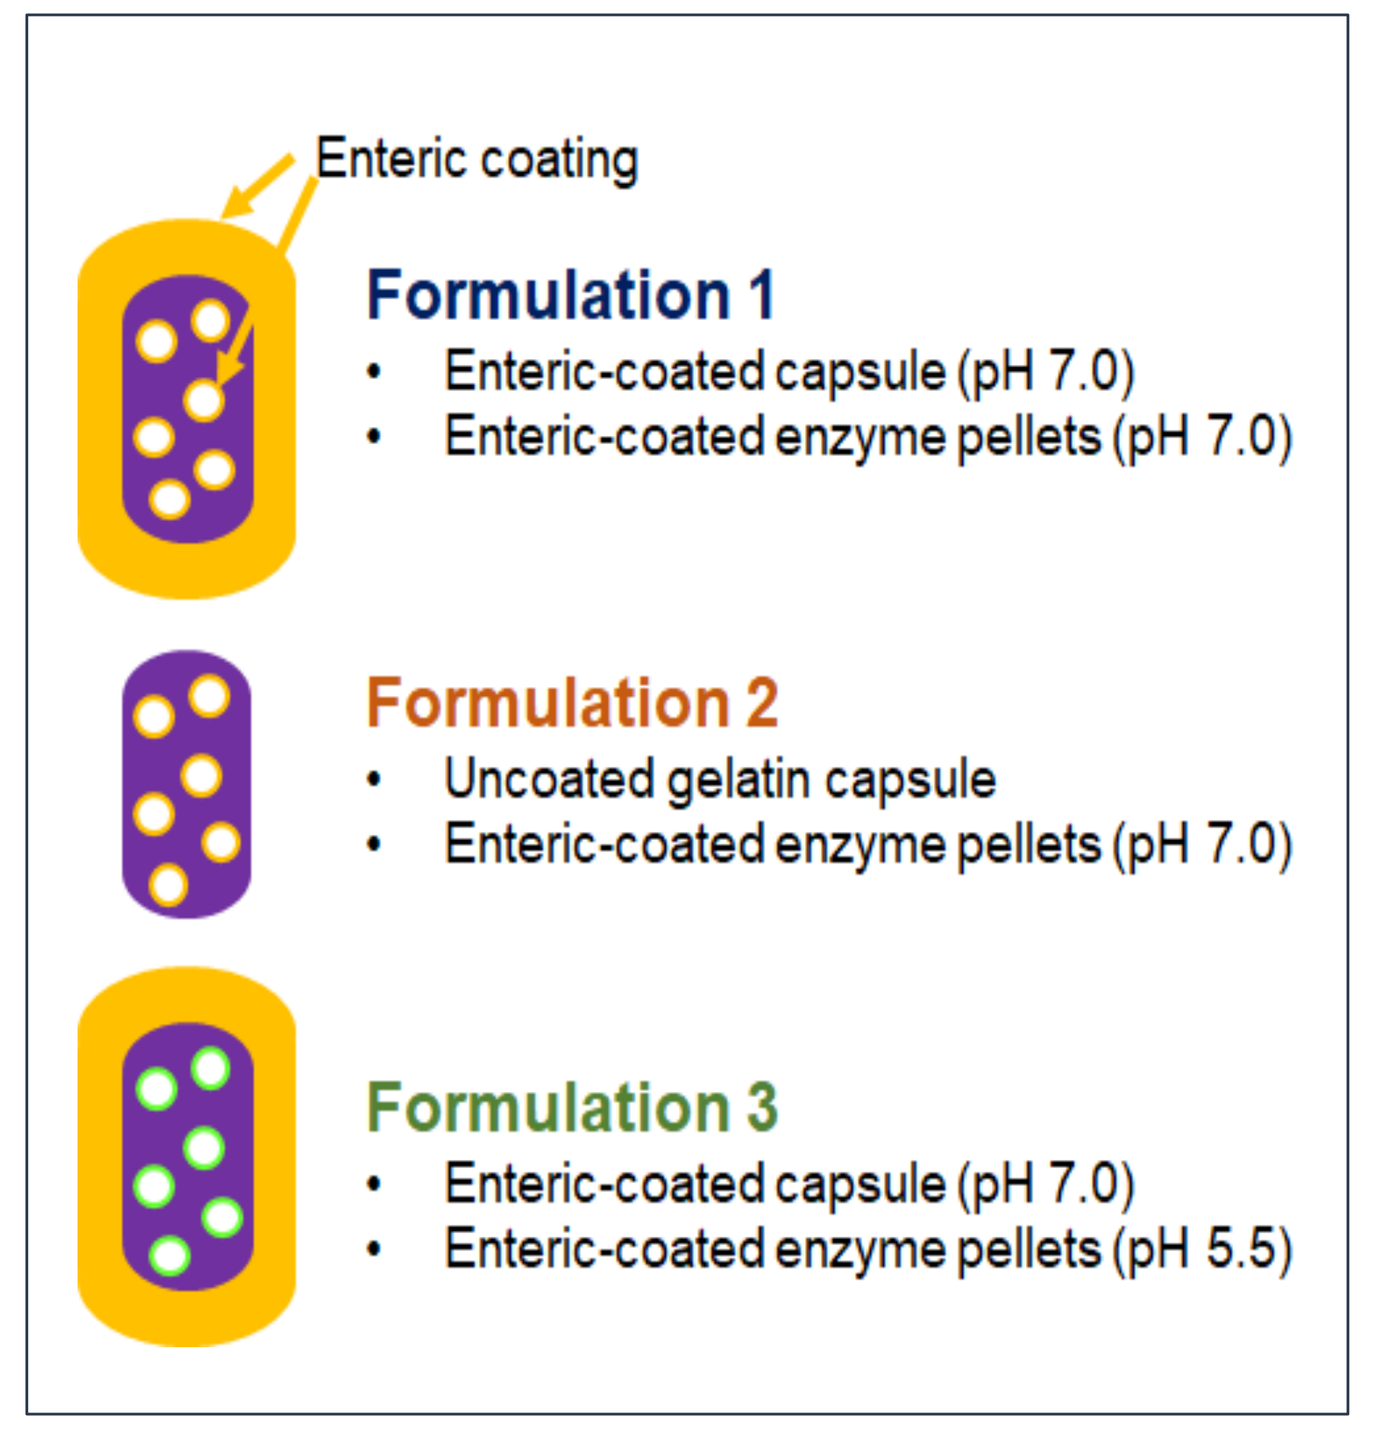
Enteric coating
Formulation 1
Enteric-coated capsule (pH 7.0)
Enteric-coated enzyme pellets (pH 7.0)
Formulation 2
Uncoated gelatin capsule
Enteric-coated enzyme pellets (pH 7.0)
Formulation 3
Enteric-coated capsule (pH 7.0)
Enteric-coated enzyme pellets (pH 5.5)

Dissolution Under Differing pH Conditions

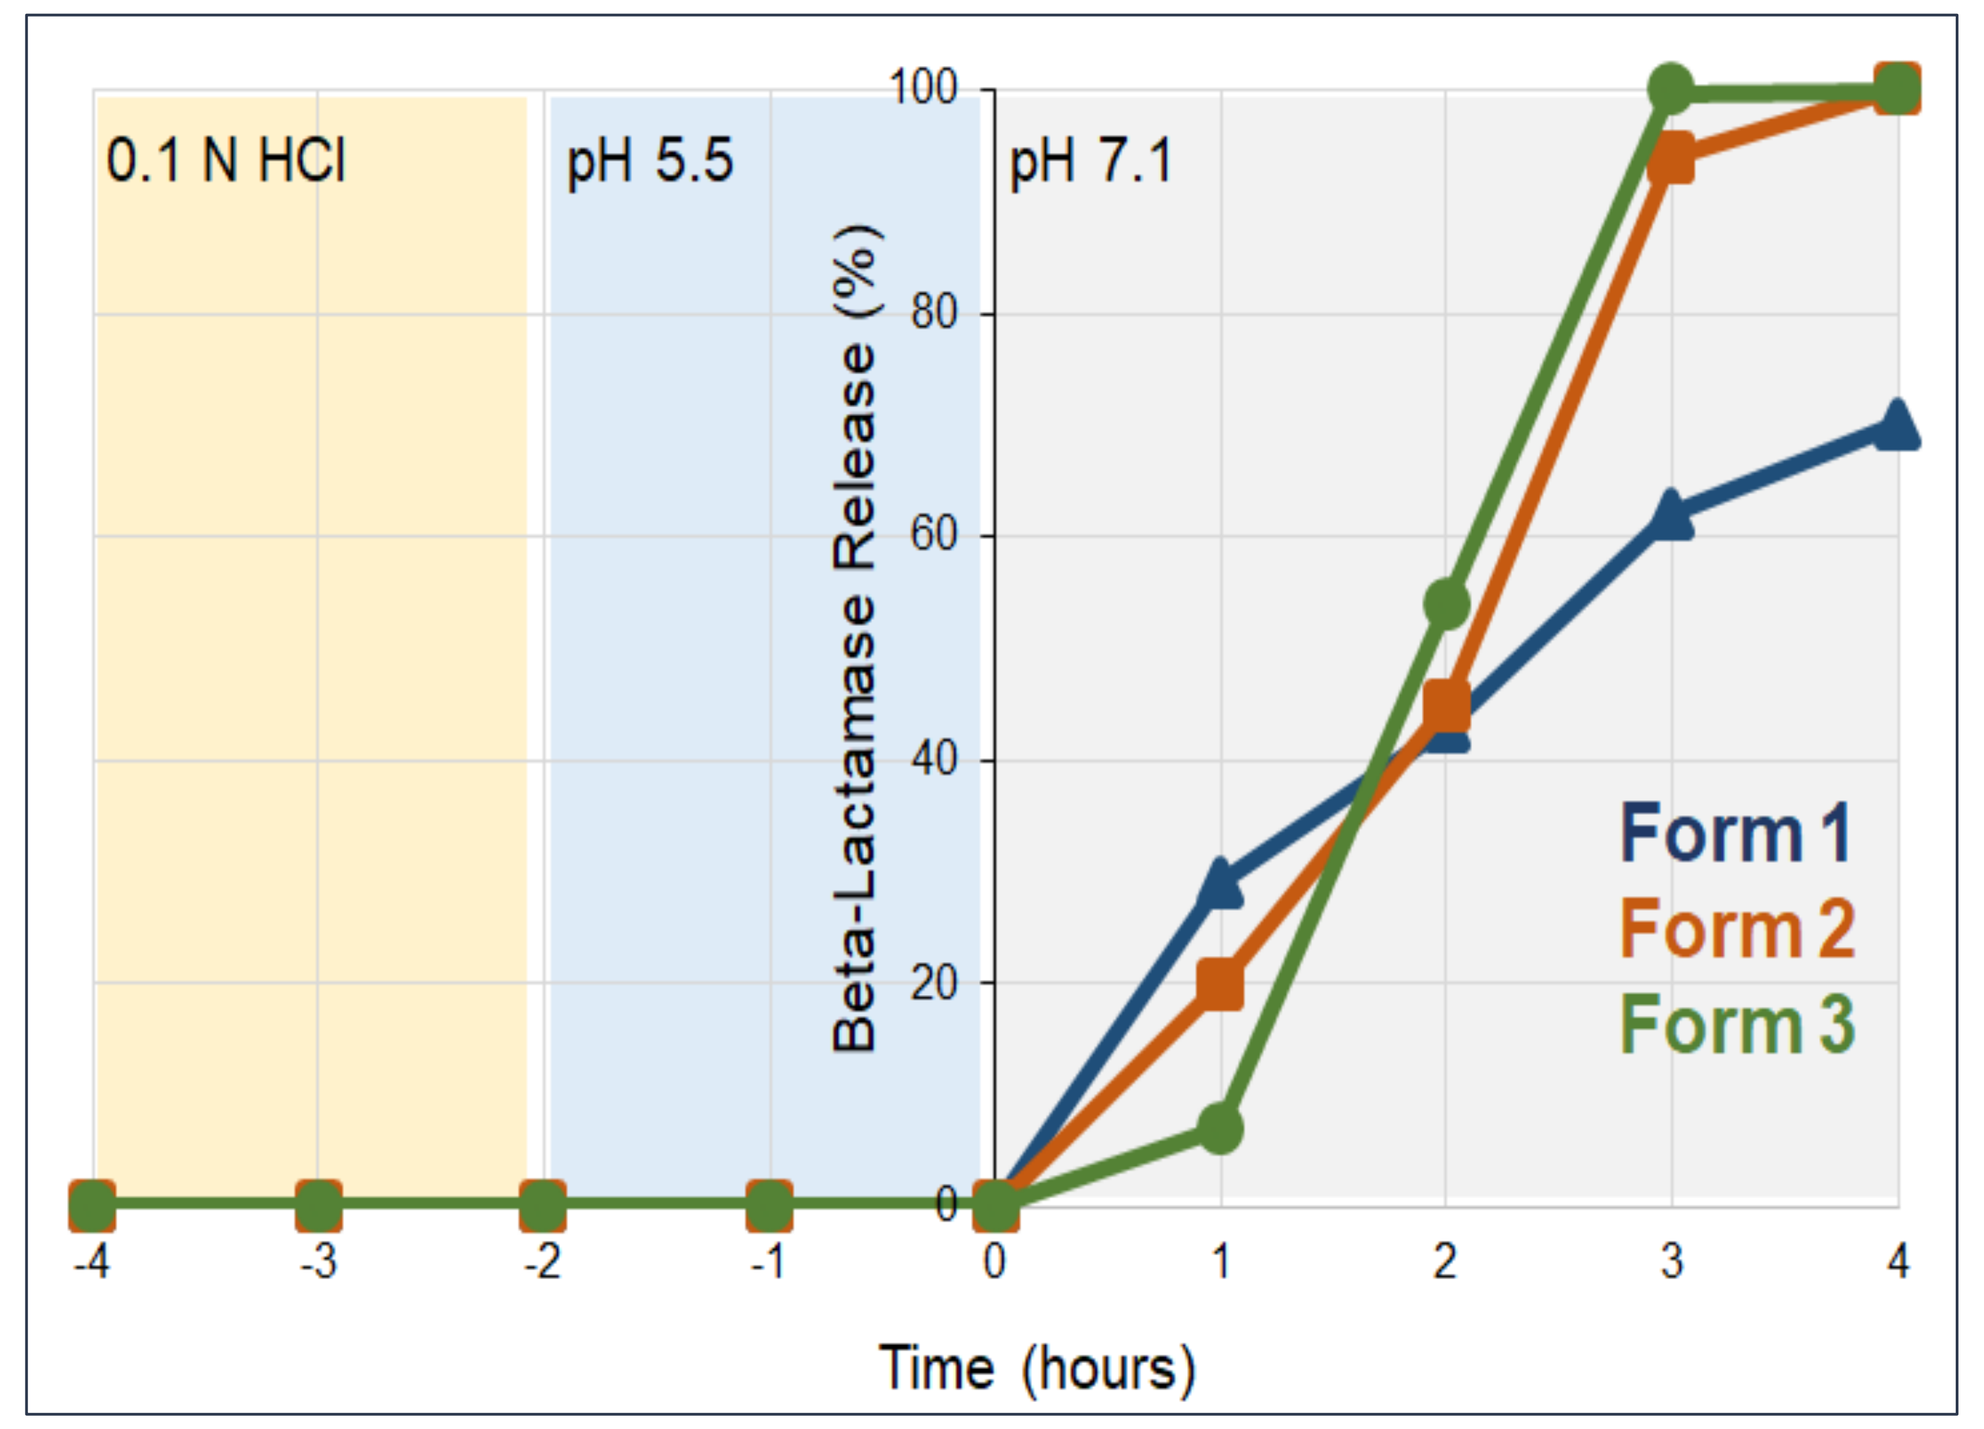
0.1 N HCl
pH 5.5
pH 7.1
Beta-Lactamase Release (%)
Time (hours)
Form 1
Form 2
Form 3

No enzyme release was detected in 0.1N HCl or at pH 5.5. At pH 7.1, dissolution occurred more slowly for Formulation 3, and by 3 hrs, was complete for Formulations 2 and 3. Formulation 1 displayed fast initial dissolution, however only 70% of total enzyme was released by 4 hrs.

In Vivo Evaluation of SYN-007 Distal-Release Formulations

The three formulations were evaluated in dogs (n=5/cohort). Animals received oral amoxicillin (80 mg/kg, TID) +/- SYN-007 formulations 1, 2, or 3 (10 mg, PO, TID) for 5 days. Serum amoxicillin levels were measured after the first and last antibiotic doses to determine if SYN-007 interfered with amoxicillin systemic absorption. For microbiome analyses, feces were collected prior to and after the last antibiotic +/- SYN-007 doses (16 doses of each total). Fecal DNA was subjected to whole genome shotgun sequence analyses using the CosmosID, Inc. metagenomics software. Microbiome analyses included assessment of bacterial species relative abundance, alpha diversity, principal coordinate analyses, and evaluations of changes to the resistome.

SYN-007 Formulation 3 Does Not Affect Amoxicillin Systemic Absorption

Blood was collected at the indicated time points after the first dose of antibiotic +/- SYN-007 (Day 1) or the last dose (Day 6) and amoxicillin serum levels were quantified.

Amoxicillin Serum Pharmacokinetics

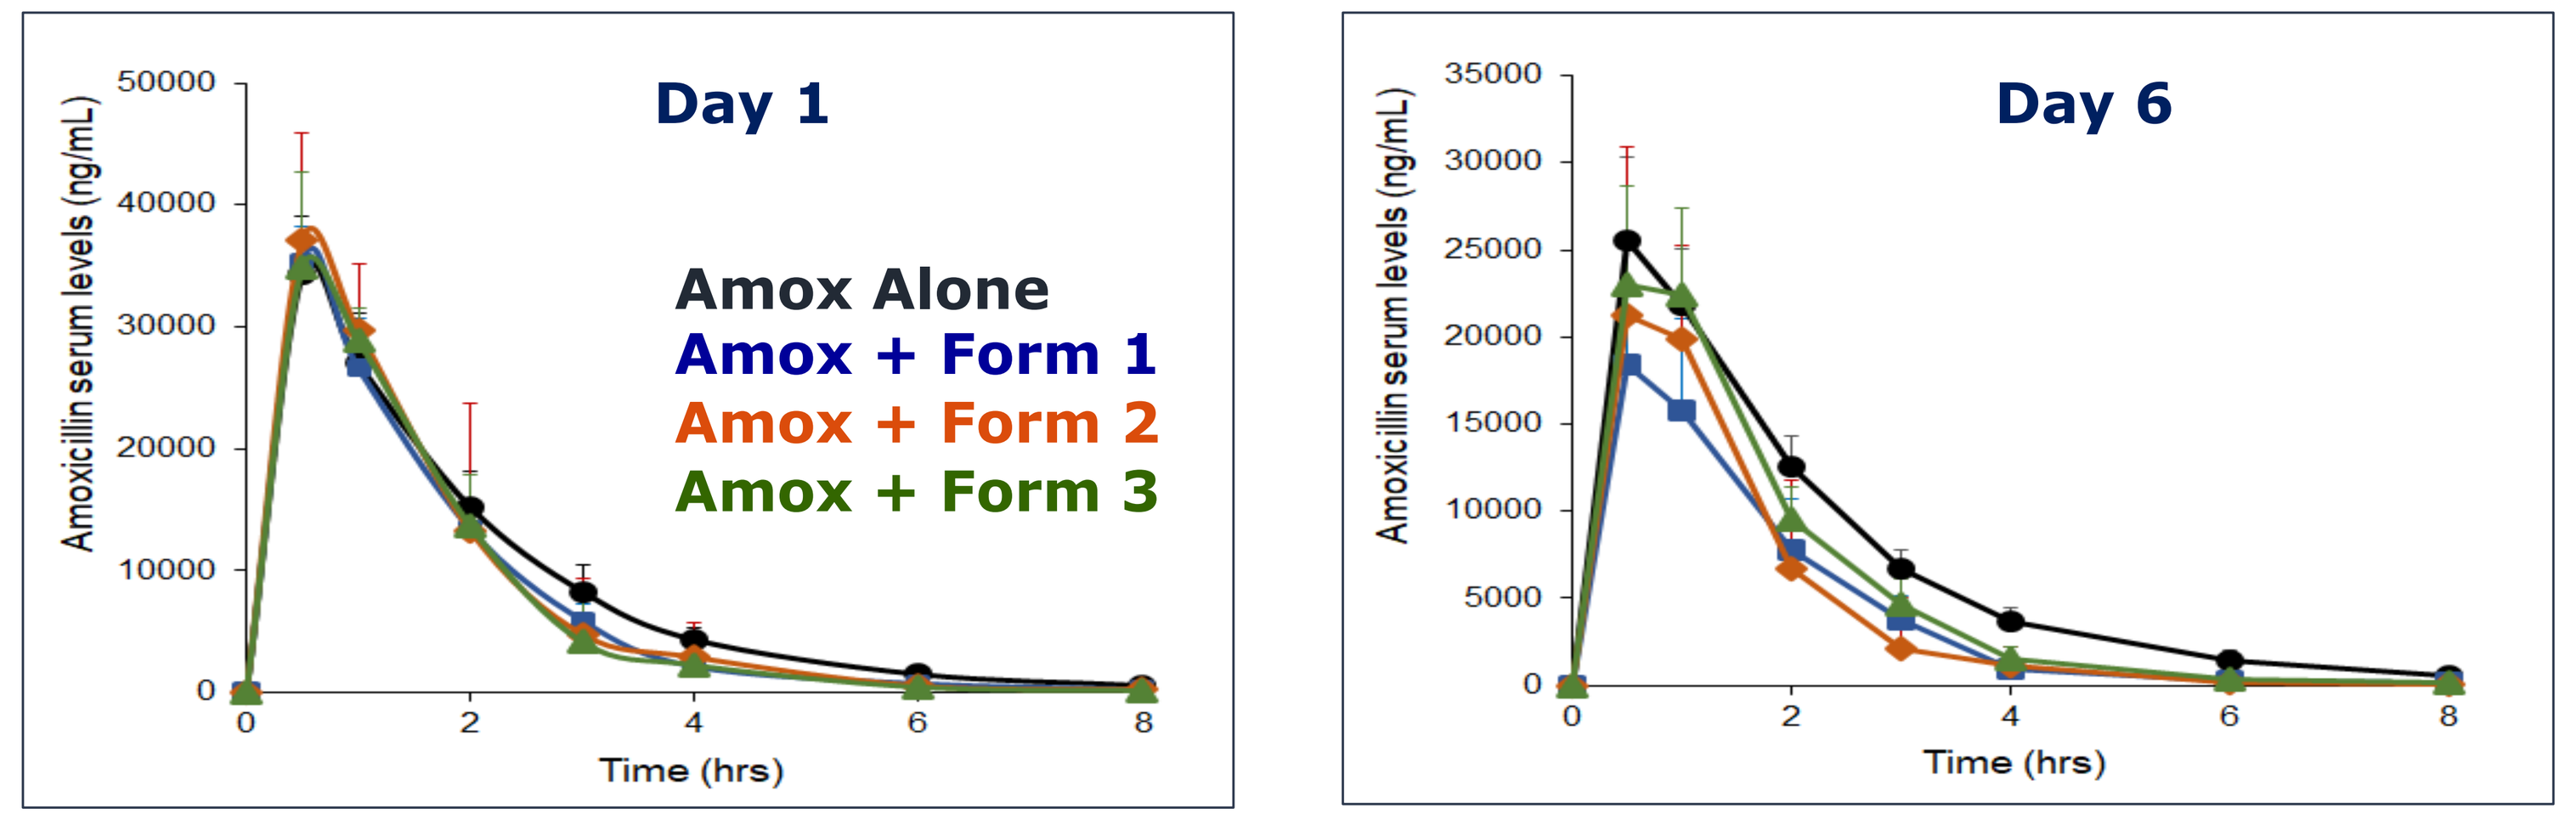
Day 1
Day 6
Amoxicillin serum levels (ng/mL)
Time (hrs)
Amox Alone
Amox + Form 1
Amox + Form 2
Amox + Form 3

Area Under the Curve

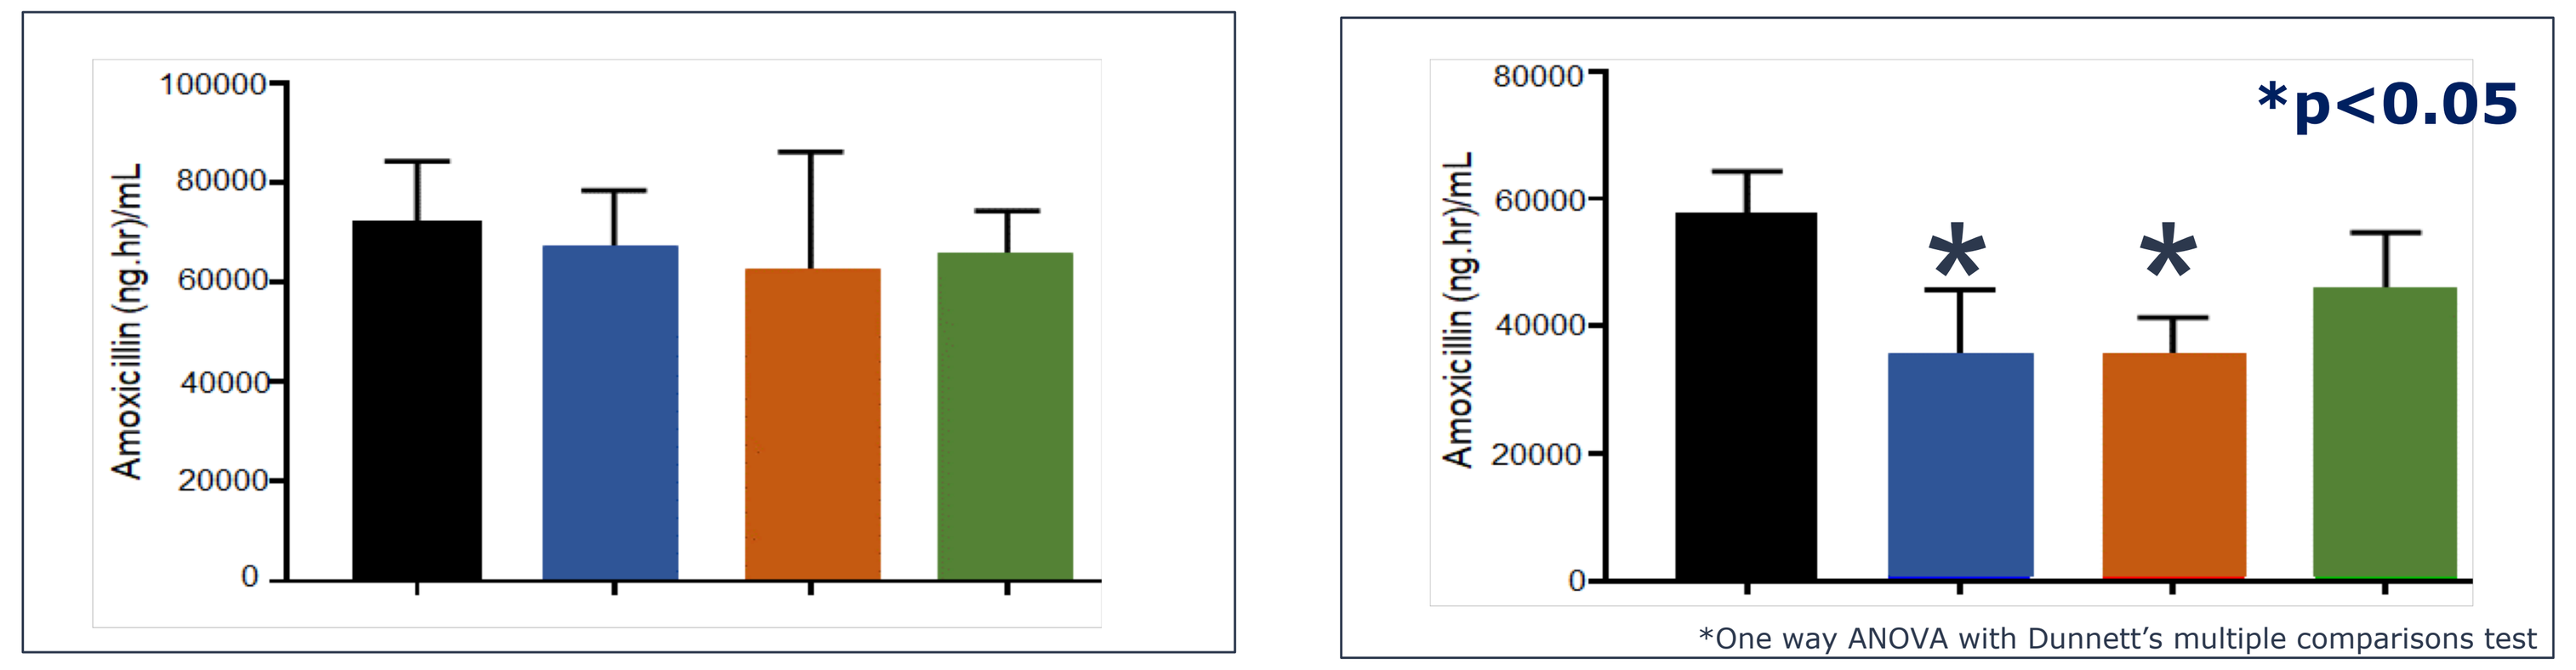
Amoxicillin (ng.hr)/mL
*p<0.05
*One way ANOVA with Dunnett's multiple comparisons test

At Day 1, amoxicillin serum levels were not significantly different in the presence or absence of each formulation of SYN-007. In contrast, at Day 6, amoxicillin serum levels were significantly reduced with SYN-007 formulations 1 (p=0.0013) and 2 (p=0.0016), but were not significantly different with formulation 3 (p=0.0980), indicating that SYN-007 formulation 3 did not interfere significantly with amoxicillin systemic absorption.

SYN-007 Protects the Gut Microbiome from Amoxicillin

Heatmaps comparing bacterial species and beta-lactamase genes present in microbiomes of dogs prior to and after amoxicillin+/- SYN-007 Formulation 3.

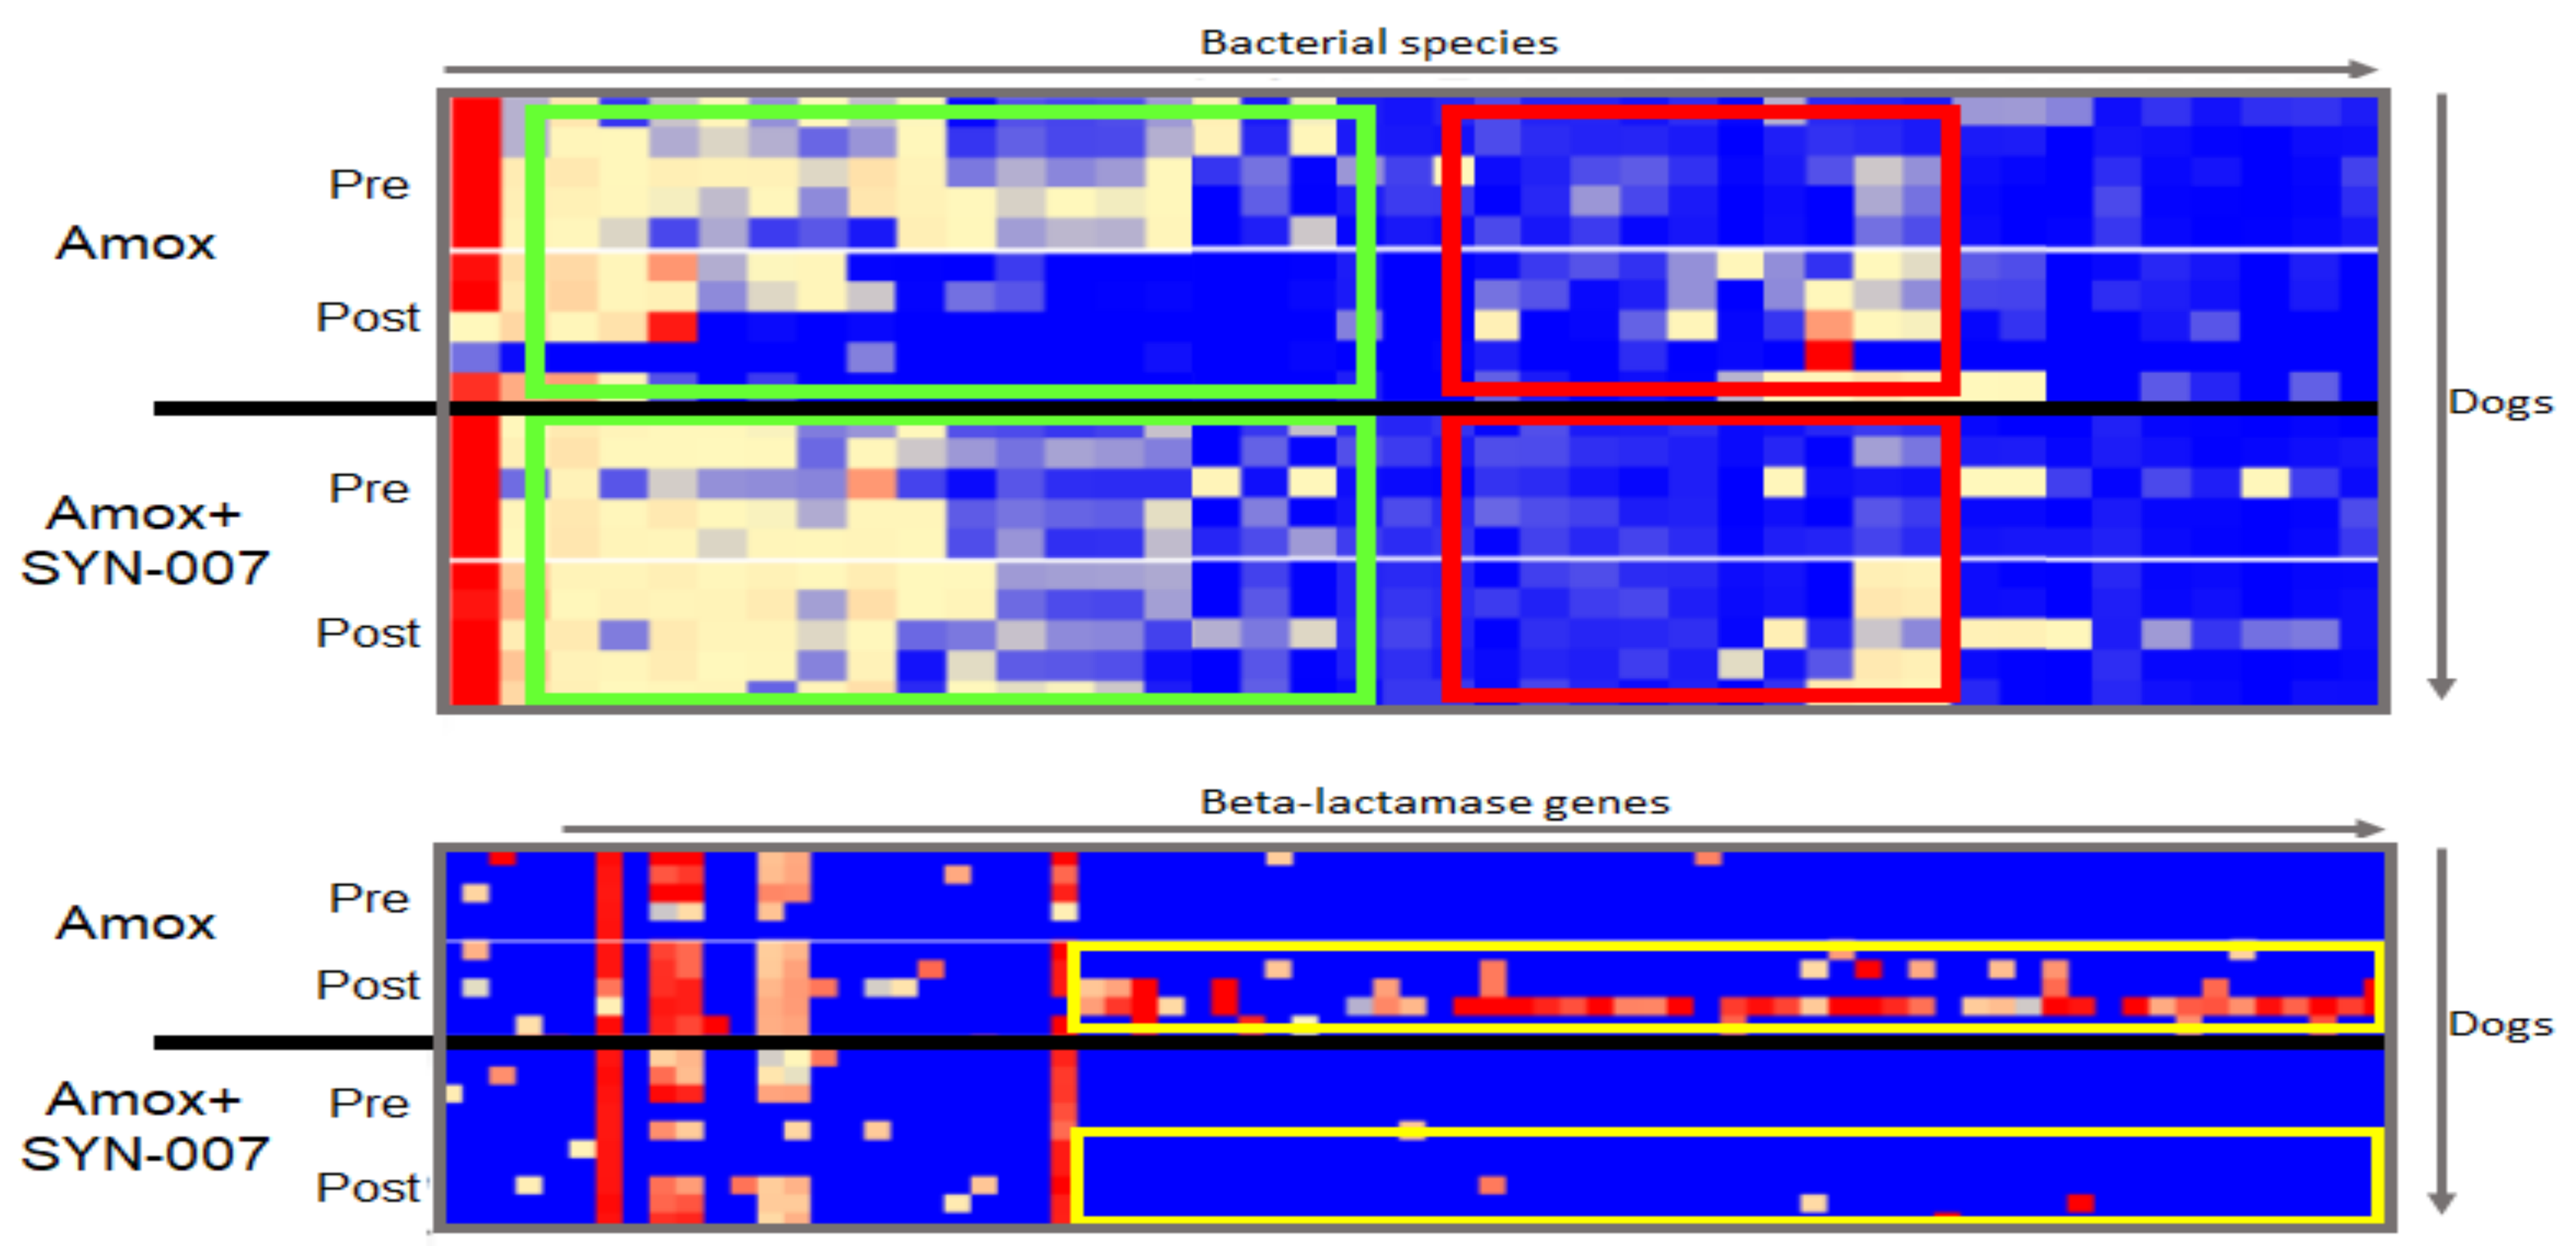
Bacterial species
Amox
Pre
Post
Amox+ SYN-007
Pre
Post
Dogs
Beta-lactamase genes
Amox
Pre
Post
Amox+ SYN-007
Pre
Post
Dogs

Amoxicillin caused depletion of some species (green boxes), overgrowth of others (red boxes), and emergence of beta-lactamase genes (yellow boxes) in addition to a broad spectrum of other resistance genes, not only those conferring resistance to beta-lactams. Microbiome changes were reduced in the presence of SYN-007.

SYN-007 Protects the Gut Microbiome from Amoxicillin

Additional microbiome analyses included Shannon Alpha Diversity and Principal Component Analyses (PCoA).

Shannon Alpha Diversity

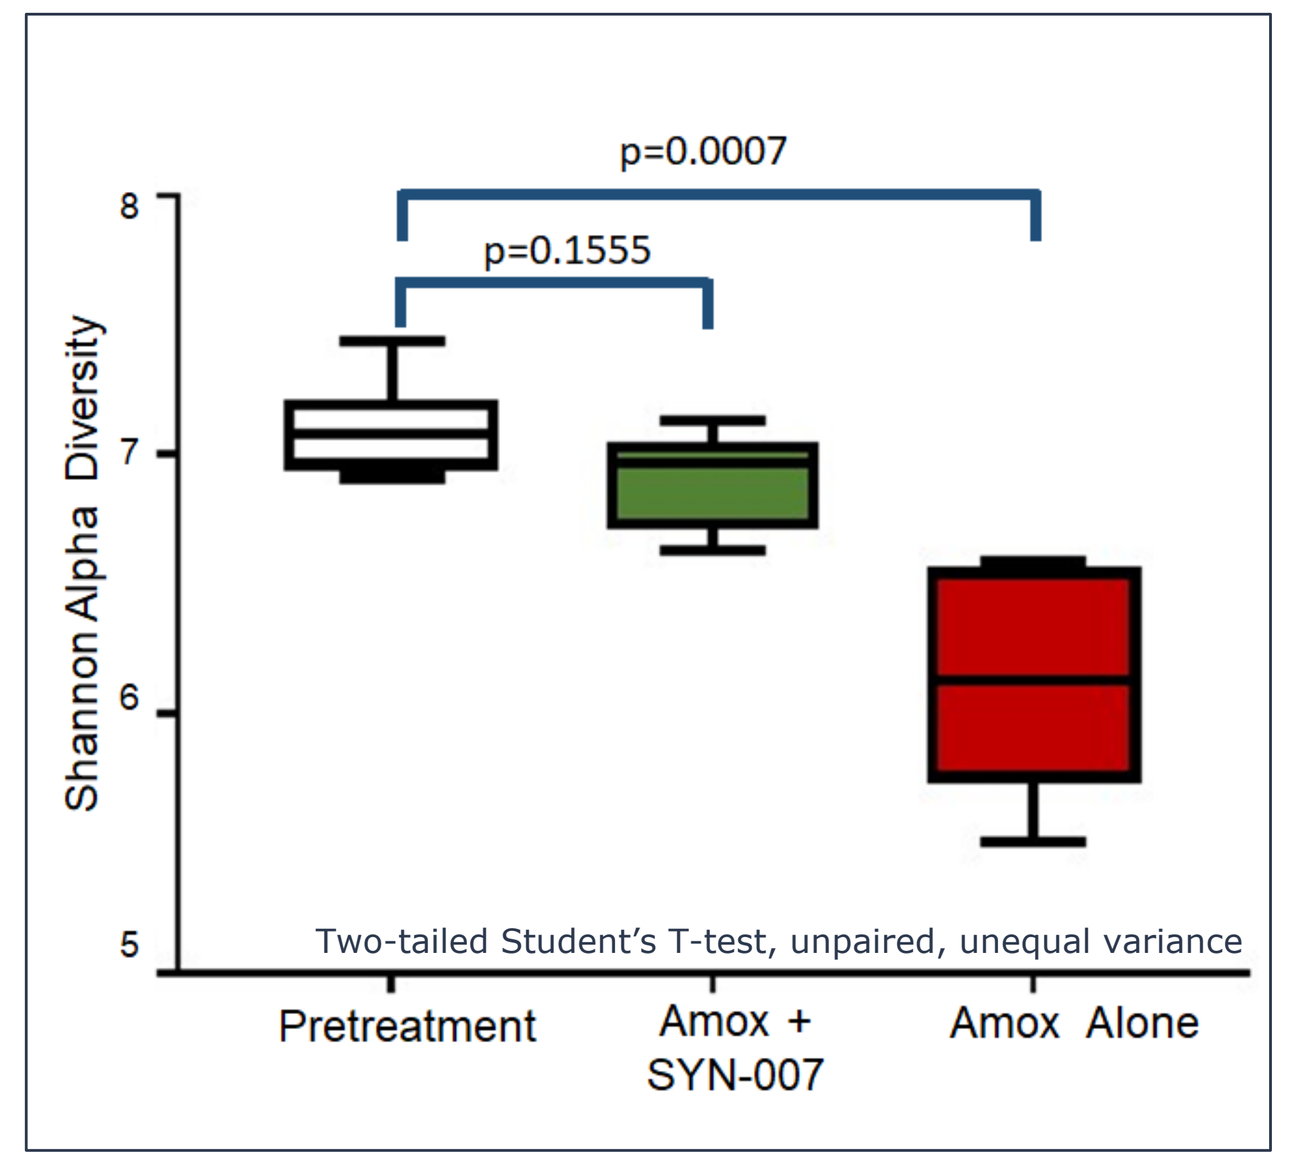
p=0.0007
p=0.1555
Shannon Alpha Diversity
Two-tailed Student's T-test, unpaired, unequal variance
Pretreatment
Amox + SYN-007
Amox Alone

Principal Component Analysis

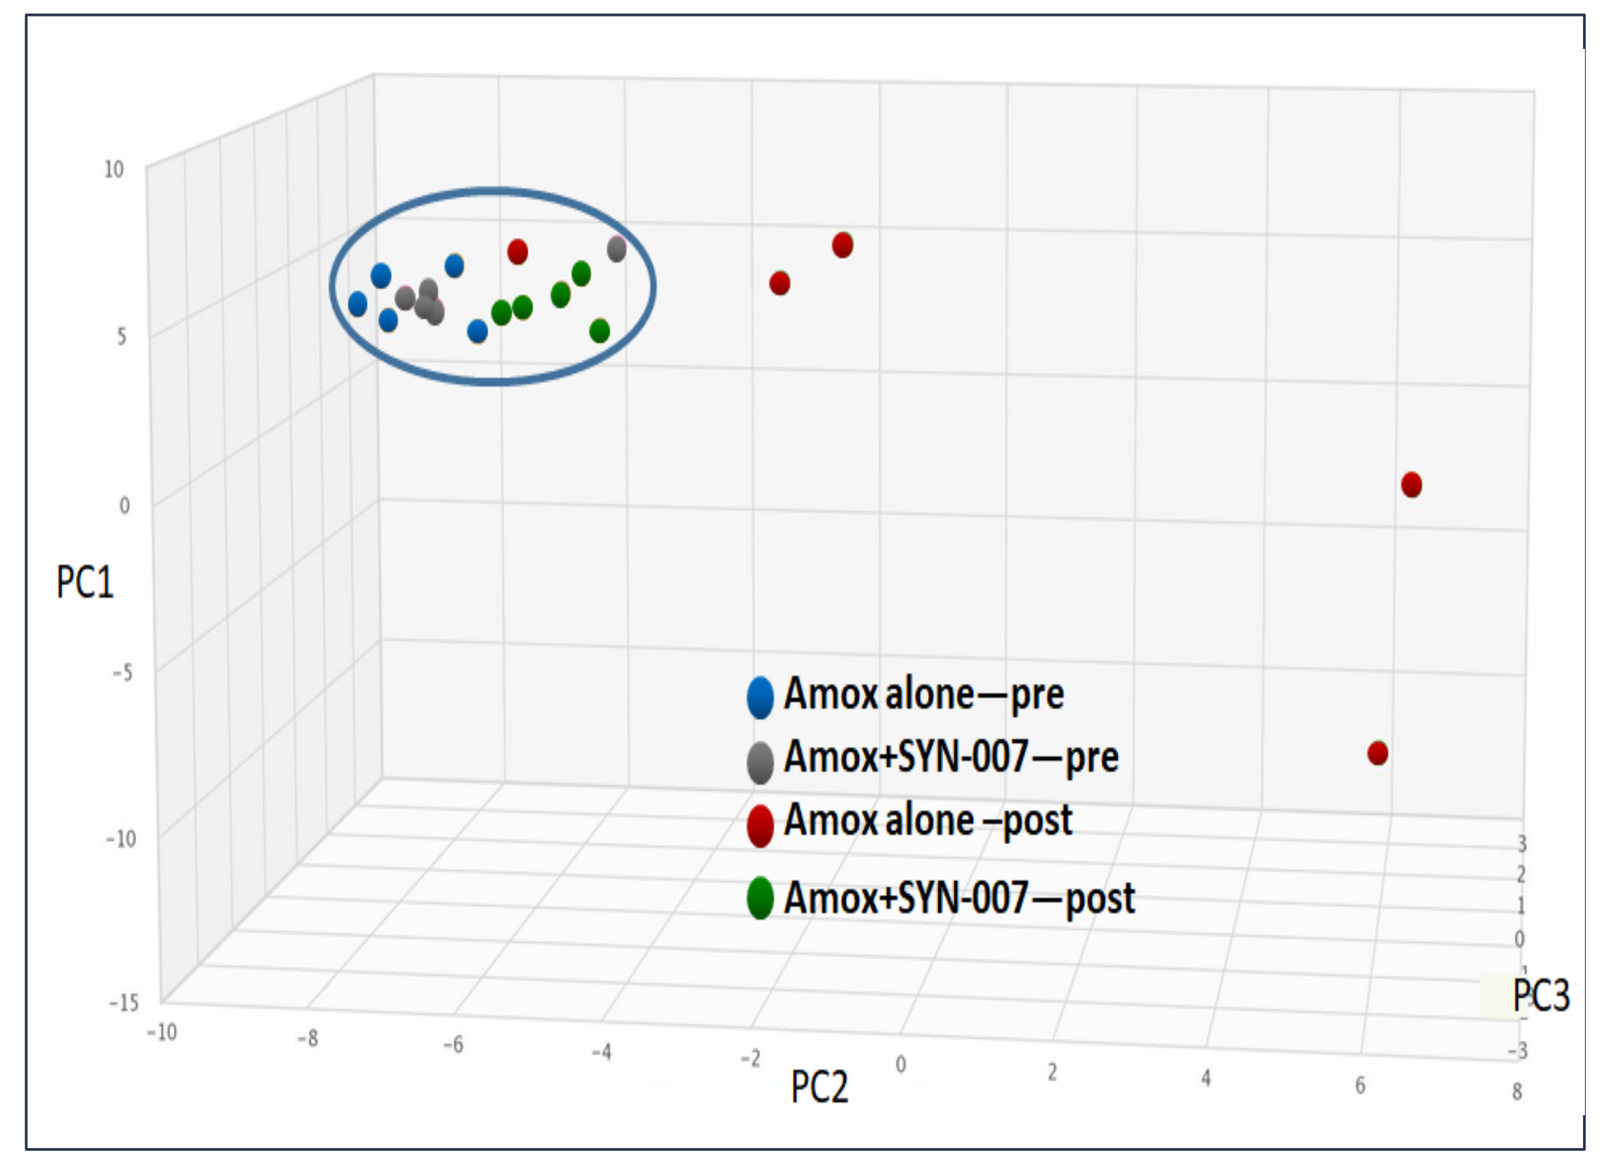
PC1
PC2
PC3
Amox alone—pre
Amox+SYN-007—pre
Amox alone –post
Amox+SYN-007—post

Compared to pretreatment microbiomes, there was no significant difference in Shannon alpha diversity in dogs treated with amoxicillin + SYN-007 (p=0.1555), while there was significant difference in the microbiomes of animals treated with amoxicillin alone (p=0.0007). Principal component analysis revealed that pretreatment microbiomes (blue and gray dots) clustered together with Amoxicillin + SYN-007 microbiomes (green dots), while 4/5 Amoxicillin alone microbiomes (red dots) fell well outside the cluster.

These data demonstrate that SYN-007 protected the gut microbiome from collateral damage caused by oral amoxicillin administration.

CONCLUSIONS

- Ribaxamase is intended as an orally-delivered beta-lactamase to protect the gut microbiome from IV beta-lactam antibiotic-mediated dysbiosis
- A phase 2b clinical study met its primary endpoint of significantly reducing *C. difficile* disease in patients receiving IV ceftriaxone + ribaxamase
- SYN-007 is a new, distal-release formulation of ribaxamase for use with oral beta-lactam antibiotics
- SYN-007 Form 3 did not interfere with oral amoxicillin absorption in dogs
- SYN-007 protected the gut microbiota from damage caused by amoxicillin in dogs
- SYN-007 reduced emergence of antibiotic resistance genes in dogs

SYN-007 has the potential to expand microbiome protection to include oral as well as IV beta-lactam antibiotics

DISCLOSURES

SC, CFF, and MK are employees of Synthetic Biologics, Inc. RRC is the founder of CosmosID, Inc., a fee-for-service provider engaged by Synthetic Biologics, Inc. BF and NAH are employees of CosmosID, Inc. This work was supported by Synthetic Biologics, Inc.

Synthetic Biologics, Inc.