Treatment of RSV Lower Respiratory Tract Infection in Two Immunocompromised Children with Polyclonal Immunoglobulin Containing Standardized Levels of Neutralizing Anti-RSV Antibodies

ER Levy[1,2], T Madigan[2], MJ Binnicker[3], J Mond[4], WC Huskins[2]

[1]Division of Pediatric Critical Care, [2]Division of Pediatric Infectious Diseases, Department of Pediatrics and [3]Division of Clinical Microbiology, Department of Laboratory Medicine and Pathology, Mayo Clinic, Rochester, MN;
[4]Chief Scientific Officer/Chief Medical Officer, ADMA Biologics, Boca Raton, FL

Background

- Respiratory syncytial virus (RSV) can cause severe lower respiratory tract infection (LRTI) in immunocompromised patients. [1,2]
- No standard, effective therapy for severe RSV LRTI currently exists.
- Use of ribavirin (inhaled or oral), pooled donor intravenous immunoglobulin (IVIG), and monoclonal anti-RSV antibodies (palivizumab) have been described. [1]
- RI-002 (ADMA Biologics) is a pooled human polyclonal IVIG containing standardized levels of neutralizing anti-RSV antibodies that is prepared via a patented process. [2,3]
- The predecessor of RI-002 was used for compassionate treatment in RSV LRTI in stem cell transplant patients. [2,3]
- RI-002 was FDA-approved in 2019 for prophylaxis in primary immunodeficiency patients.

Methods

- Two children with T-cell lymphoblastic lymphoma and neutropenia secondary to chemotherapy were included
- Both patients had RSV LRTI
- Both received PO ribavirin and IVIG
- Both were treated with RI-002 under an emergency FDA Investigational New Drug application

RI-002 [3,4]

Table 2. Comparison of respiratory virus titers of RI-002 and commercial IVIG batches [11].

Virus	Ratio of geometric means (95% CI) (RI-002/commercial IVIG)^a	p-Value^b
RSV	1.861 (1.249, 2.771)	0.003
PIV 1	1.792 (1.282, 2.505)	0.001
OC43	1.610 (1.127, 2.301)	0.010
PIV 2	1.601 (1.160,2.210)	0.005
229E	1.494 (1.144, 1.950)	0.004
Flu A	1.402 (1.067, 1.843)	0.016
Flu B	1.316 (1.026, 1.688)	0.031
hMPV	1.264 (0.990, 1.613)	0.060
PIV 1 and 2	1.694 (1.250, 2.296)	0.001
OC43 and 229E	1.551 (1.237, 1.945)	<0.001
All viruses^c	1.529 (1.227, 1.907)	<0.001

[3]Wasserman RL *et al.* 2017. PMID: 29035131

- Pooled polyclonal intravenous immune globulin with standardized levels of neutralizing anti-RSV antibodies
- Derived from human plasma donors tested to have high levels of neutralizing antibodies to RSV

Patient #1: Clinical Course

- Admitted (Hospital Day [HD] #1) with fever, neutropenia, and nasal congestion; NP RSV PCR positive
- HD#9-17: Progressive respiratory failure
- HD#17: Intubated for respiratory failure. Initiated on ribavirin PO 30 mg/kg/day, received 0.5 g/kg IVIG and palivizumab
- HD#18: High frequency oscillation, paralysis, and nitric oxide. Mediastinal air leak. Emergent IND FDA approval for RI-002
- HD#20: RI-002 1st Dose (1.5 g/kg)
- HD#22: RI-002 2nd Dose (0.75 g/kg)
- HD#23: Progressive air leak, elective V-V ECMO
- HD#30: Chest CT severe ground glass opacities with new diffuse bronchiectasis; down-trending crossing point (Cp) values
- HD#34: RI-002 3rd Dose (0.75 g/kg)
- HD#38-42: Improved pulmonary compliance, negative ETT RSV PCR so ribavirin discontinued; on dexamethasone; Tracheostomy HD#43
- HD#52: V-V ECMO discontinued, weaning respiratory support

Discharged home HD#88

1 month later returned for tracheostomy de-cannulation, currently at respiratory baseline with no support

Patient 1: Serial CT images at level of left atrium while on RI-002

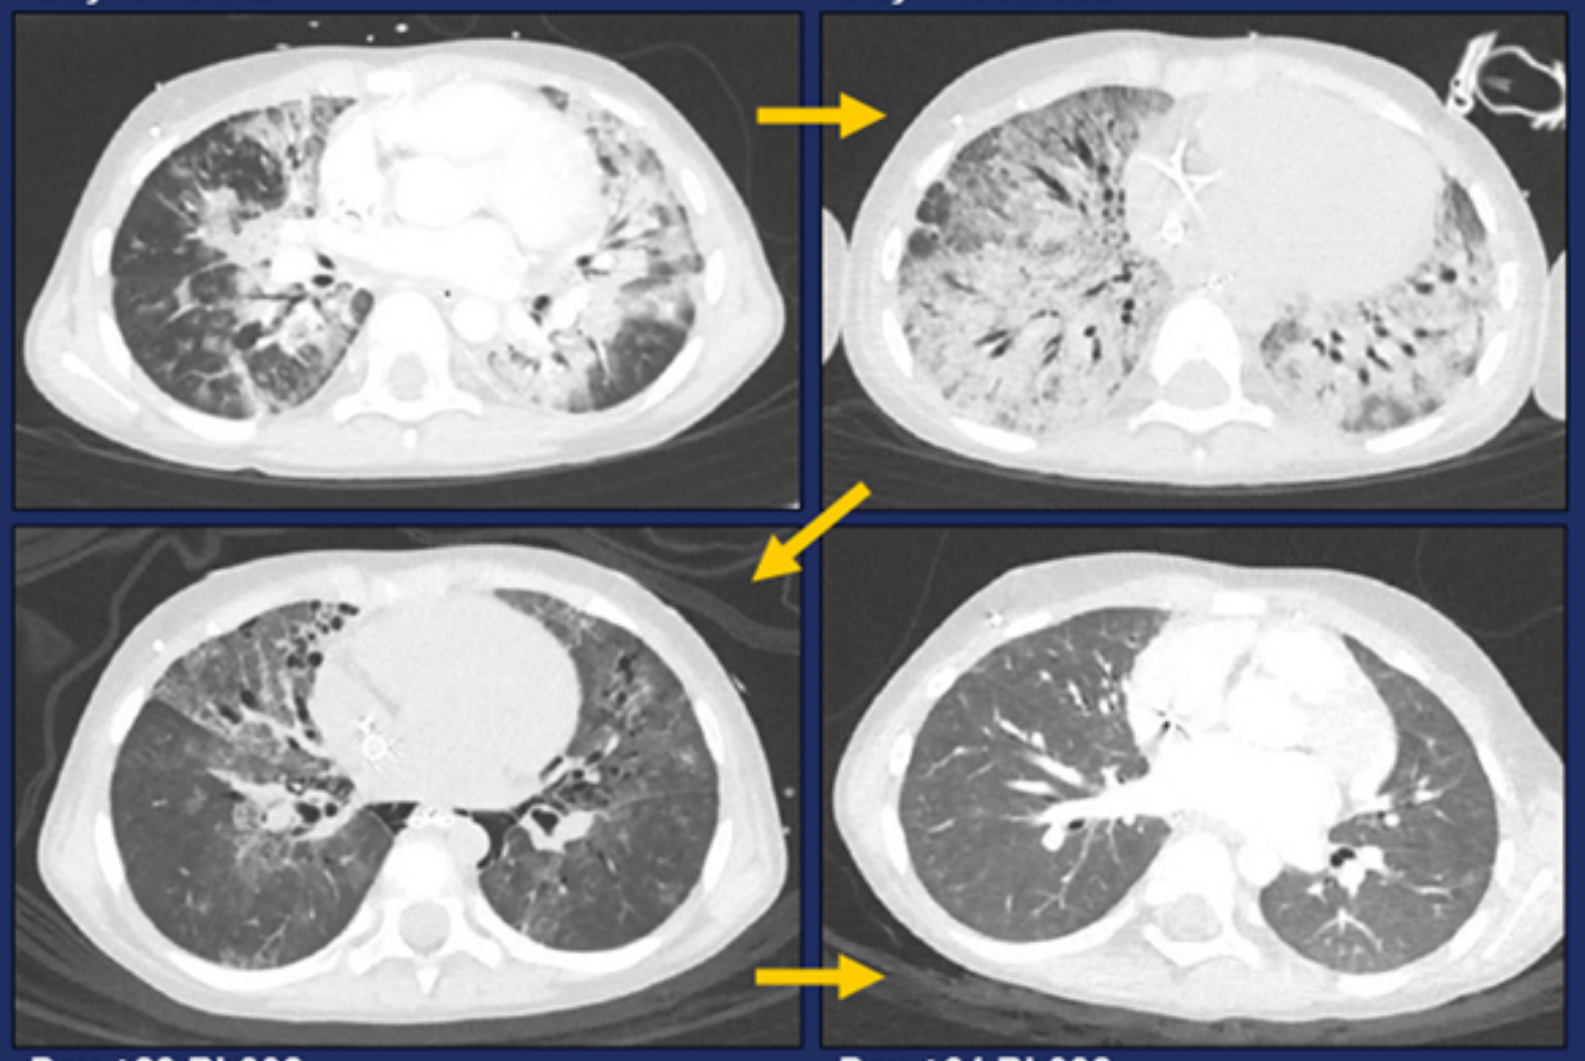
Day -3 RI-002
Day +10 RI-002
Day +23 RI-002
Day +64 RI-002

Patient #1 Microbiology Data

Hospital Day (HD)	RI-002 treatment	Specimen type	RSV PCR Result	RSV PCR Cp*	Viral Culture
HD5		NP swab	Positive	n/a	n/a
HD10		BAL	Positive	21.1	RSV positive
HD17		BAL	Positive	23.2	RSV positive
HD20	1.5g/kg				
HD22	0.75g/kg				
HD24		BAL	Positive	29	No growth
HD29		NP swab	Negative	n/a	n/a
HD29		ETT aspirate	Positive	33.8	No growth
HD33		ETT aspirate	Positive	35.7	No growth
HD34	0.75g/kg				
HD36		ETT aspirate	Negative	n/a	No growth
HD37		NP swab	Negative	n/a	n/a

*PCR Cp values are a semi-quantitative determination of strength of positivity
NP, nasopharyngeal; BAL, bronchoalveolar lavage;
ETT, endotracheal; n/a, not available; Cp, crossing point

Patient #2: Clinical Course

- Admitted (HD#1) with fever, neutropenia, nasal congestion, cough, and stridor; NP RSV PCR positive
- HD#1: 1L NC (2/19)
- HD#2-3: IVIG 0.5 g/kg + Oral ribavirin 30 mg/kg/day
- HD#4: Dexamethasone x 3 days for stridor
- HD#5: RI-002 (1.5 g/kg)
- HD#6: Off oxygen, afebrile
- HD#7: RI-002 (0.75g/kg), ribavirin discontinued

Discharged home HD#8

Conclusions

- Polyclonal immunoglobulin (RI-002) contains high levels of neutralizing anti-RSV antibodies.
- In immunocompromised children:
 - RI-002 may be useful in the treatment of severe RSV LRTI.
 - RI-002 may be useful in preventing RSV infection or its progression to severe LRTI.
- Future studies on the role of RI-002 in the treatment and prevention of RSV LRTI in immunocompromised children are warranted.

References

1. Alansari K, Toaimah FH, Almatar DH, El Tatawy LA, Davidson BL, Qusad MIM. Monoclonal Antibody Treatment of RSV Bronchiolitis in Young Infants: A Randomized Trial. Pediatrics. 2019 Mar;143(3). pii: e20182308. doi: 10.1542/peds.2018-2308. Epub 2019 Feb 13. PubMed PMID: 30760509.
2. Falsey AR, Koval C, DeVincenzo JP, Walsh EE. Compassionate use experience with high-titer respiratory syncytical virus (RSV) immunoglobulin in RSV-infected immunocompromised persons. Transpl Infect Dis. 2017 Apr;19(2). doi: 10.1111/tid.12657. Epub 2017 Mar 6. PubMed PMID: 28054734.
3. Wasserman RL, Greener BN, Mond J. RI-002, an intravenous immunoglobulin containing high titer neutralizing antibody to RSV and other respiratory viruses for use in primary immunodeficiency disease and other immune compromised populations. Expert Rev Clin Immunol. 2017 Dec;13(12):1107-1119. doi: 10.1080/1744666X.2017.1389647. Epub 2017 Oct 16. Review. PubMed PMID: 29035131.
4. Wasserman RL, Lumry W, Harris J 3rd, Levy R, Stein M, Forbes L, Cunningham-Rundles C, Melamed I, Kobayashi AL, Du W, Kobayashi R. Efficacy, Safety, and Pharmacokinetics of a New 10 % Liquid Intravenous Immunoglobulin Containing High Titer Neutralizing Antibody to RSV and Other Respiratory Viruses in Subjects with Primary Immunodeficiency Disease. J Clin Immunol. 2016 Aug;36(6):590-9. doi: 10.1007/s10875-016-0308-z. Epub 2016 Jun 20. PubMed PMID: 27324887; PubMed Central PMCID: PMC4940435.
5. https://www.admabiologics.com/ (accessed 4/22/2019)

Mayo Clinic Children's Center