P2327

Antibacterial activity of ibezapolstat against antimicrobial resistant clinical strains of *Clostridioides difficile*

Eugénie Bassères, Taryn Eubank, Khurshida Begum, M. Jahangir Alam, Jinhee Jo, Thanh M. Le, Chris K. Lancaster, Anne J Gonzales-Luna, Kevin W. Garey

University of Houston College of Pharmacy, Houston, TX USA

College of Pharmacy
UNIVERSITY OF HOUSTON

BACKGROUND

- Although novel therapeutics are in development for CDI, including the recent approval of live biotherapeutic products, only three antibiotics have made it to phase III trials since the approval of FDX in 2011, and none have filed for Food and Drug Administration approval.
- Antimicrobial resistance is emerging in clinical strains of *Clostridioides difficile*.
- New antibiotics and particularly those with activity against multidrug-resistant (MDR) strains are urgently needed.
- Ibezapolstat (IBZ), is a DNA polymerase IIIC inhibitor currently in phase II clinical trials.
- IBZ has potent *in vitro* activity against wild-type, susceptible strains but its effect on *C. difficile* strains with reduced susceptibility to metronidazole (MTZ), vancomycin (VAN), or fidaxomicin (FDX) has not been tested.

METHODS

In vitro activity, bactericidal, and time kill activity of IBZ versus comparators was evaluated against 100 clinical strains tested of which 59 had reduced susceptibility to other *C. difficile* antibiotics.

- **Antibiotics**: IBZ, FDX, VAN, MTZ
- **MIC** testing by agar dilution according to CLSI standard for anaerobic bacteria.
- **ATP-bioluminescence assay** (BacTiter-Glo assay, Promega Corp.) Signal decays calculated according to the formula:

%relative luminescence=
(well luminescence-mean of media background)/(mean of growth control-mean of media background)x100%

- **Time-kill kinetics**:
 - bactericidal activity: reduction of $\geq 3\log_{10}$ in viability relative to the starting inoculum after 24h exposure to antibiotics
 - Bacteriostatic: $<3\log_{10}$ killing compared to starting inoculum
- **Light microscopy**:
 - reference strain R20291
 - Clinical strains MT4883 (reduced susceptibility to VAN) and FDXR28 (reduced susceptibility to FDX)

OBJECTIVE

Test the antibacterial properties of ibezapolstat against multidrug resistant *C. difficile* strains.

RESULTS

Ibezapolstat bactericidal activity against multidrug resistant *C. difficile* strains

		Ibezapolstat			
	N	MIC_{50}	MIC_{90}	ATP_{50}	ATP_{90}
Ribotype					
All	100	4	8	4	16
F027	27	4	8	4	8
F106	13	8	16	4	32
F014-020	12	4	8	4	8
F002	7	8	8	4	16
Other	41	4	8	4	32
Susceptibility					
VAN susceptible	61	8	8	4	32
VAN non-susceptible	39	4	8	4	8
FDX susceptible	61	4	8	4	16
FDX non-susceptible	39	4	8	4	32
MTZ susceptible	66	8	8	4	32
MTZ non-susceptible	34	4	8	4	8
No. of antibiotics[a] non-susceptible					
0	41	4	8	4	32
1	26	8	8	4	32
2	13	4	8	4	8
3	20	4	4	2	8

a Antibiotics: VAN, FDX, and MTZ.

Ibezapolstat bactericidal activity against multidrug resistant *C. difficile* strains

		Drug concentration (μg/mL)					
Test strain	Drug	0	4	8	16	32	64
CD630	IBZ	.	−2.22	−2.82	−2.22	−2.4	−2.75
	FDX	.	−3.52	−3.57	−3.57	−3.14	−4.01
	VAN	.	1.45	−0.08	−0.68	−1.72	−2.52
	Control	2.92	.	.	.	.	.
R20291	IBZ	.	−1.61	−1.94	−3.43	−3.94	−2.74
	FDX	.	−3.53	−4.25	−4.45	−1.92	−2.7
	VAN	.	2.53	−1.18	−2.61	−2.13	−2.1
	Control	2.27	.	.	.	.	.
FDXR28	IBZ	.	−0.68	−1.9	−0.98	−0.77	−1.04
	FDX	.	2.29	2.56	2.56	2.64	1.26
	Control	2.55	.	.	.	.	.
MT5094	IBZ	.	−3.6	−3.73	−2.92	−3.13	−3.55
	VAN	.	3.09	2.74	2.77	1.52	−1.44
	Control	2.72	.	.	.	.	.

a Results represent the change in $\log_{10}$ CFU/mL relative to 0 h (initial concentration). CD630 and R20291 are laboratory *C. difficile* strains. FDXR28 is a fidaxomicin non-susceptible isolate, and MT5094 is a vancomycin non-susceptible isolate.

Time-kill profiles for IBZ and comparators against reference and clinical *C. difficile* strains with reduced susceptibility

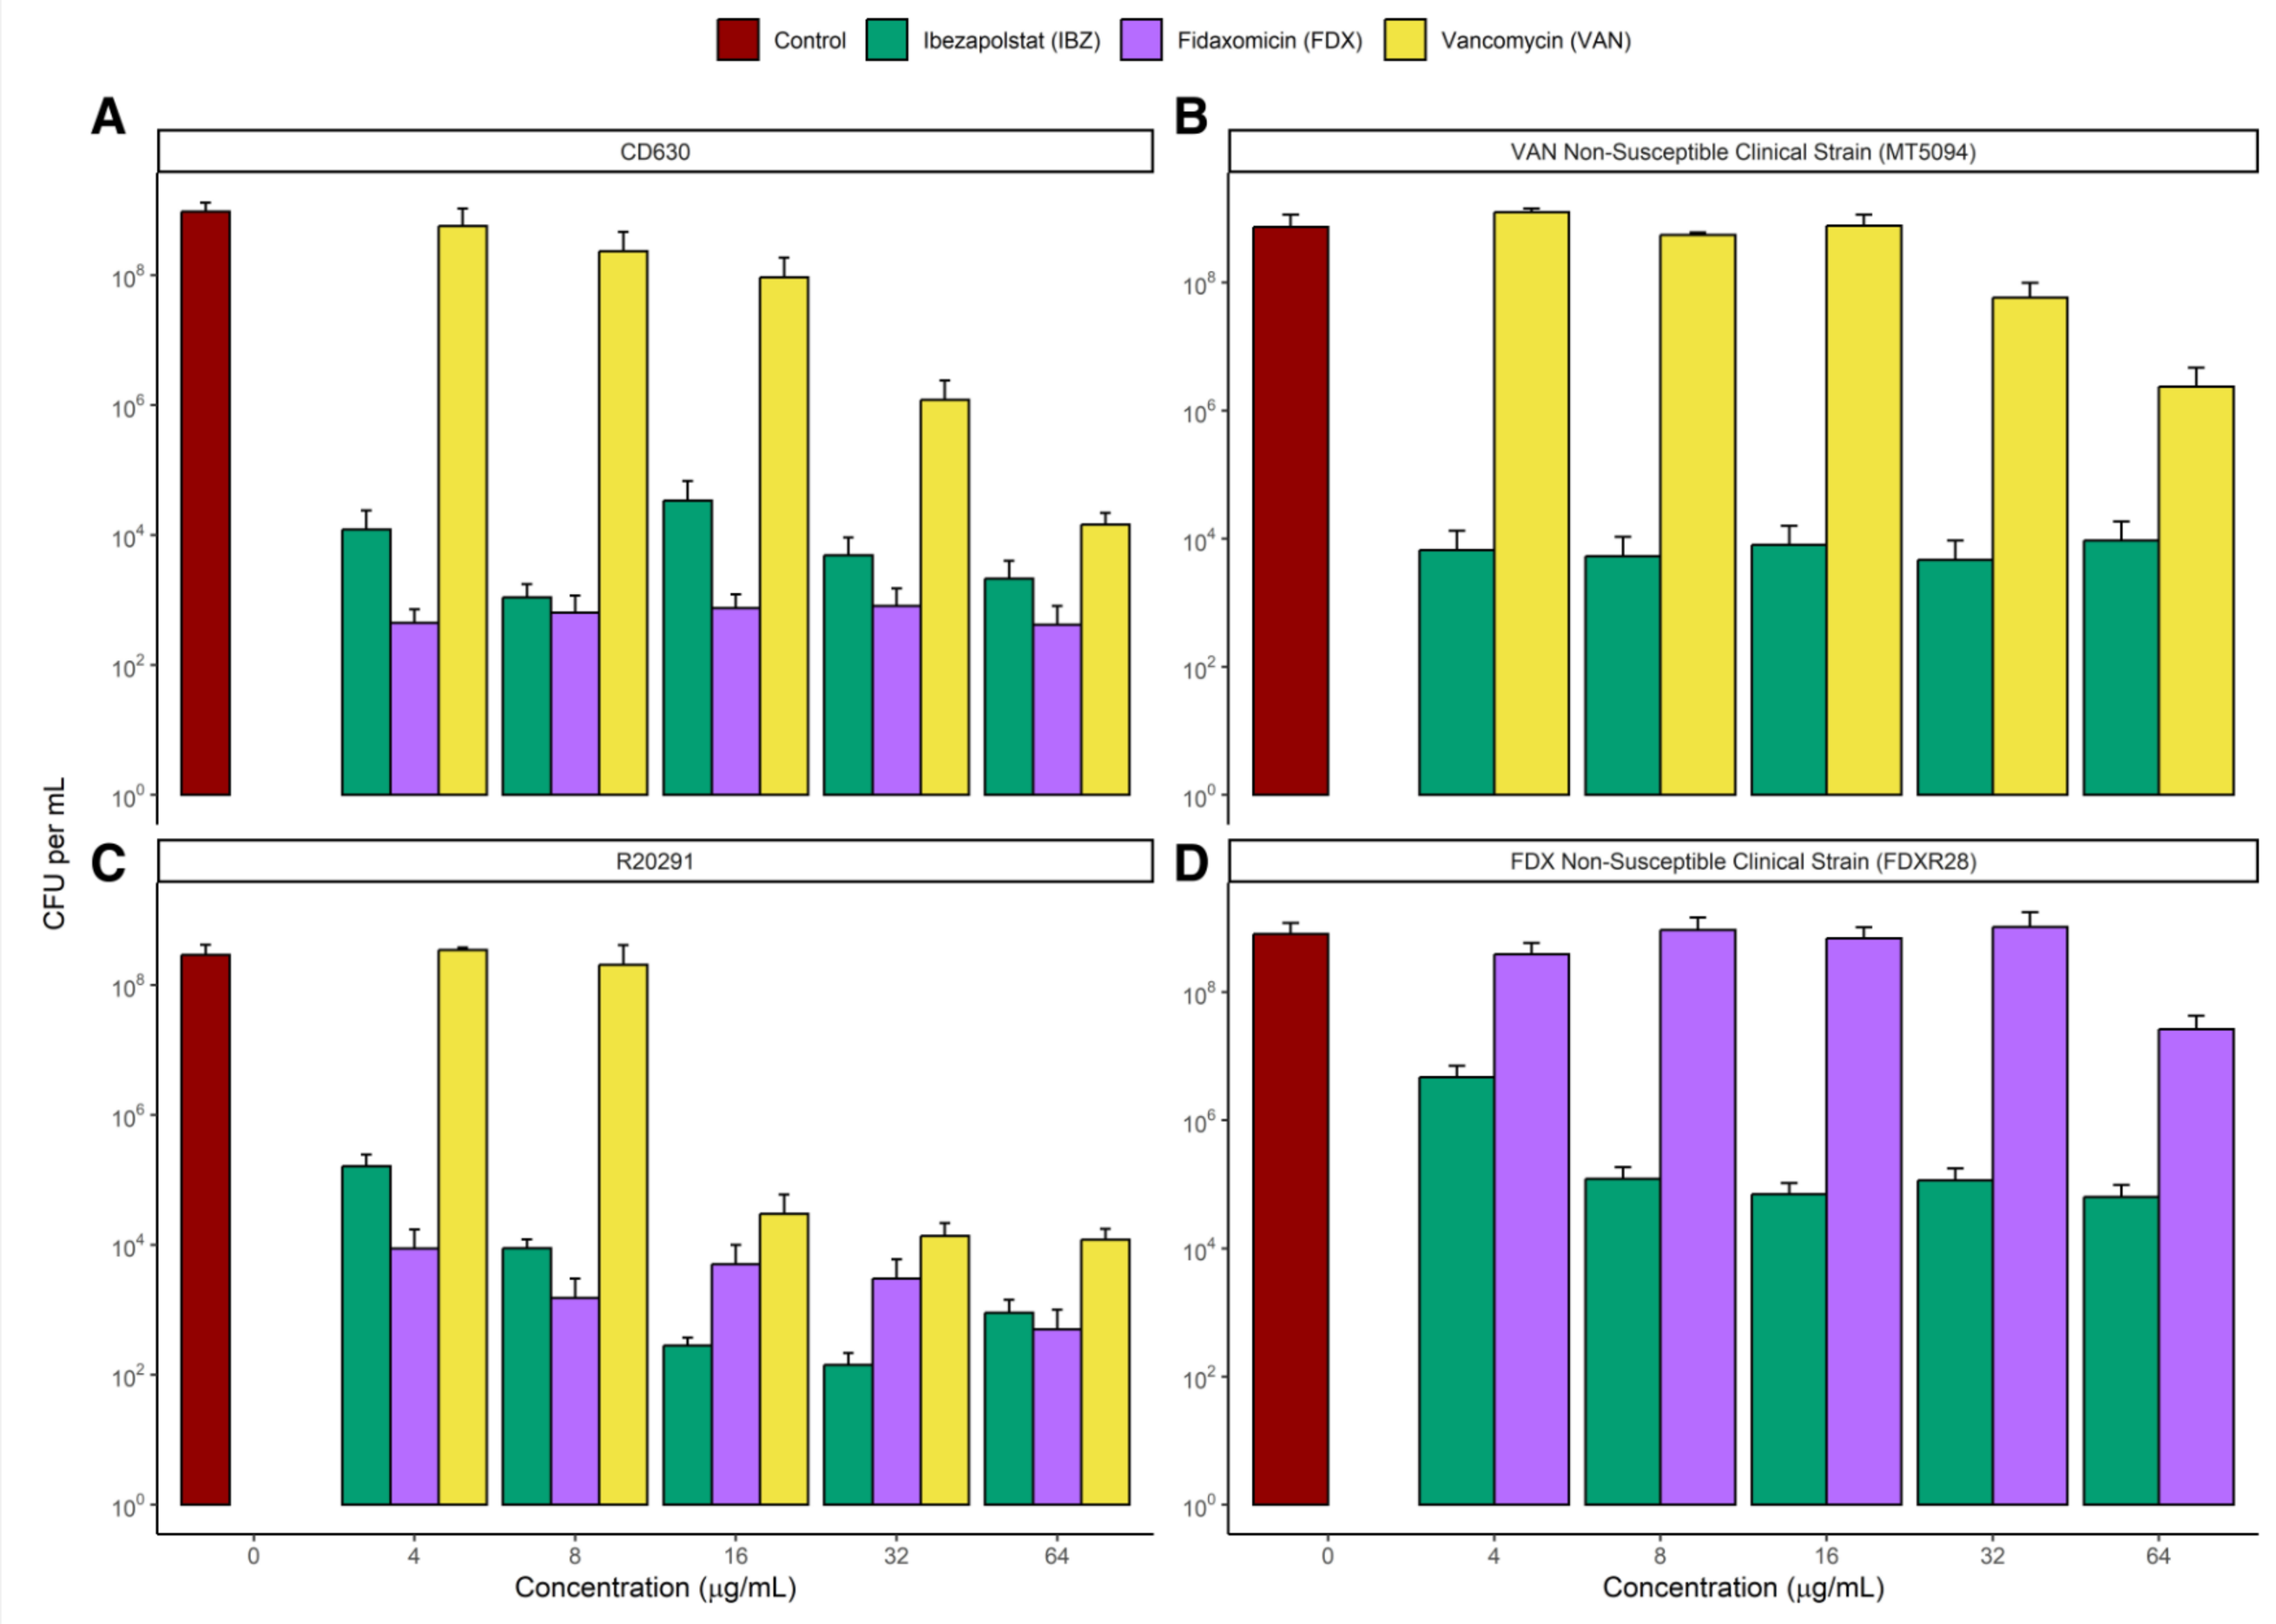
Control
Ibezapolstat (IBZ)
Fidaxomicin (FDX)
Vancomycin (VAN)
A
CD630
B
VAN Non-Susceptible Clinical Strain (MT5094)
C
R20291
D
FDX Non-Susceptible Clinical Strain (FDXR28)
CFU per mL
Concentration (μg/mL)

Ibezapolstat pharmacologic activity against reference and clinical strains of *C. difficile*

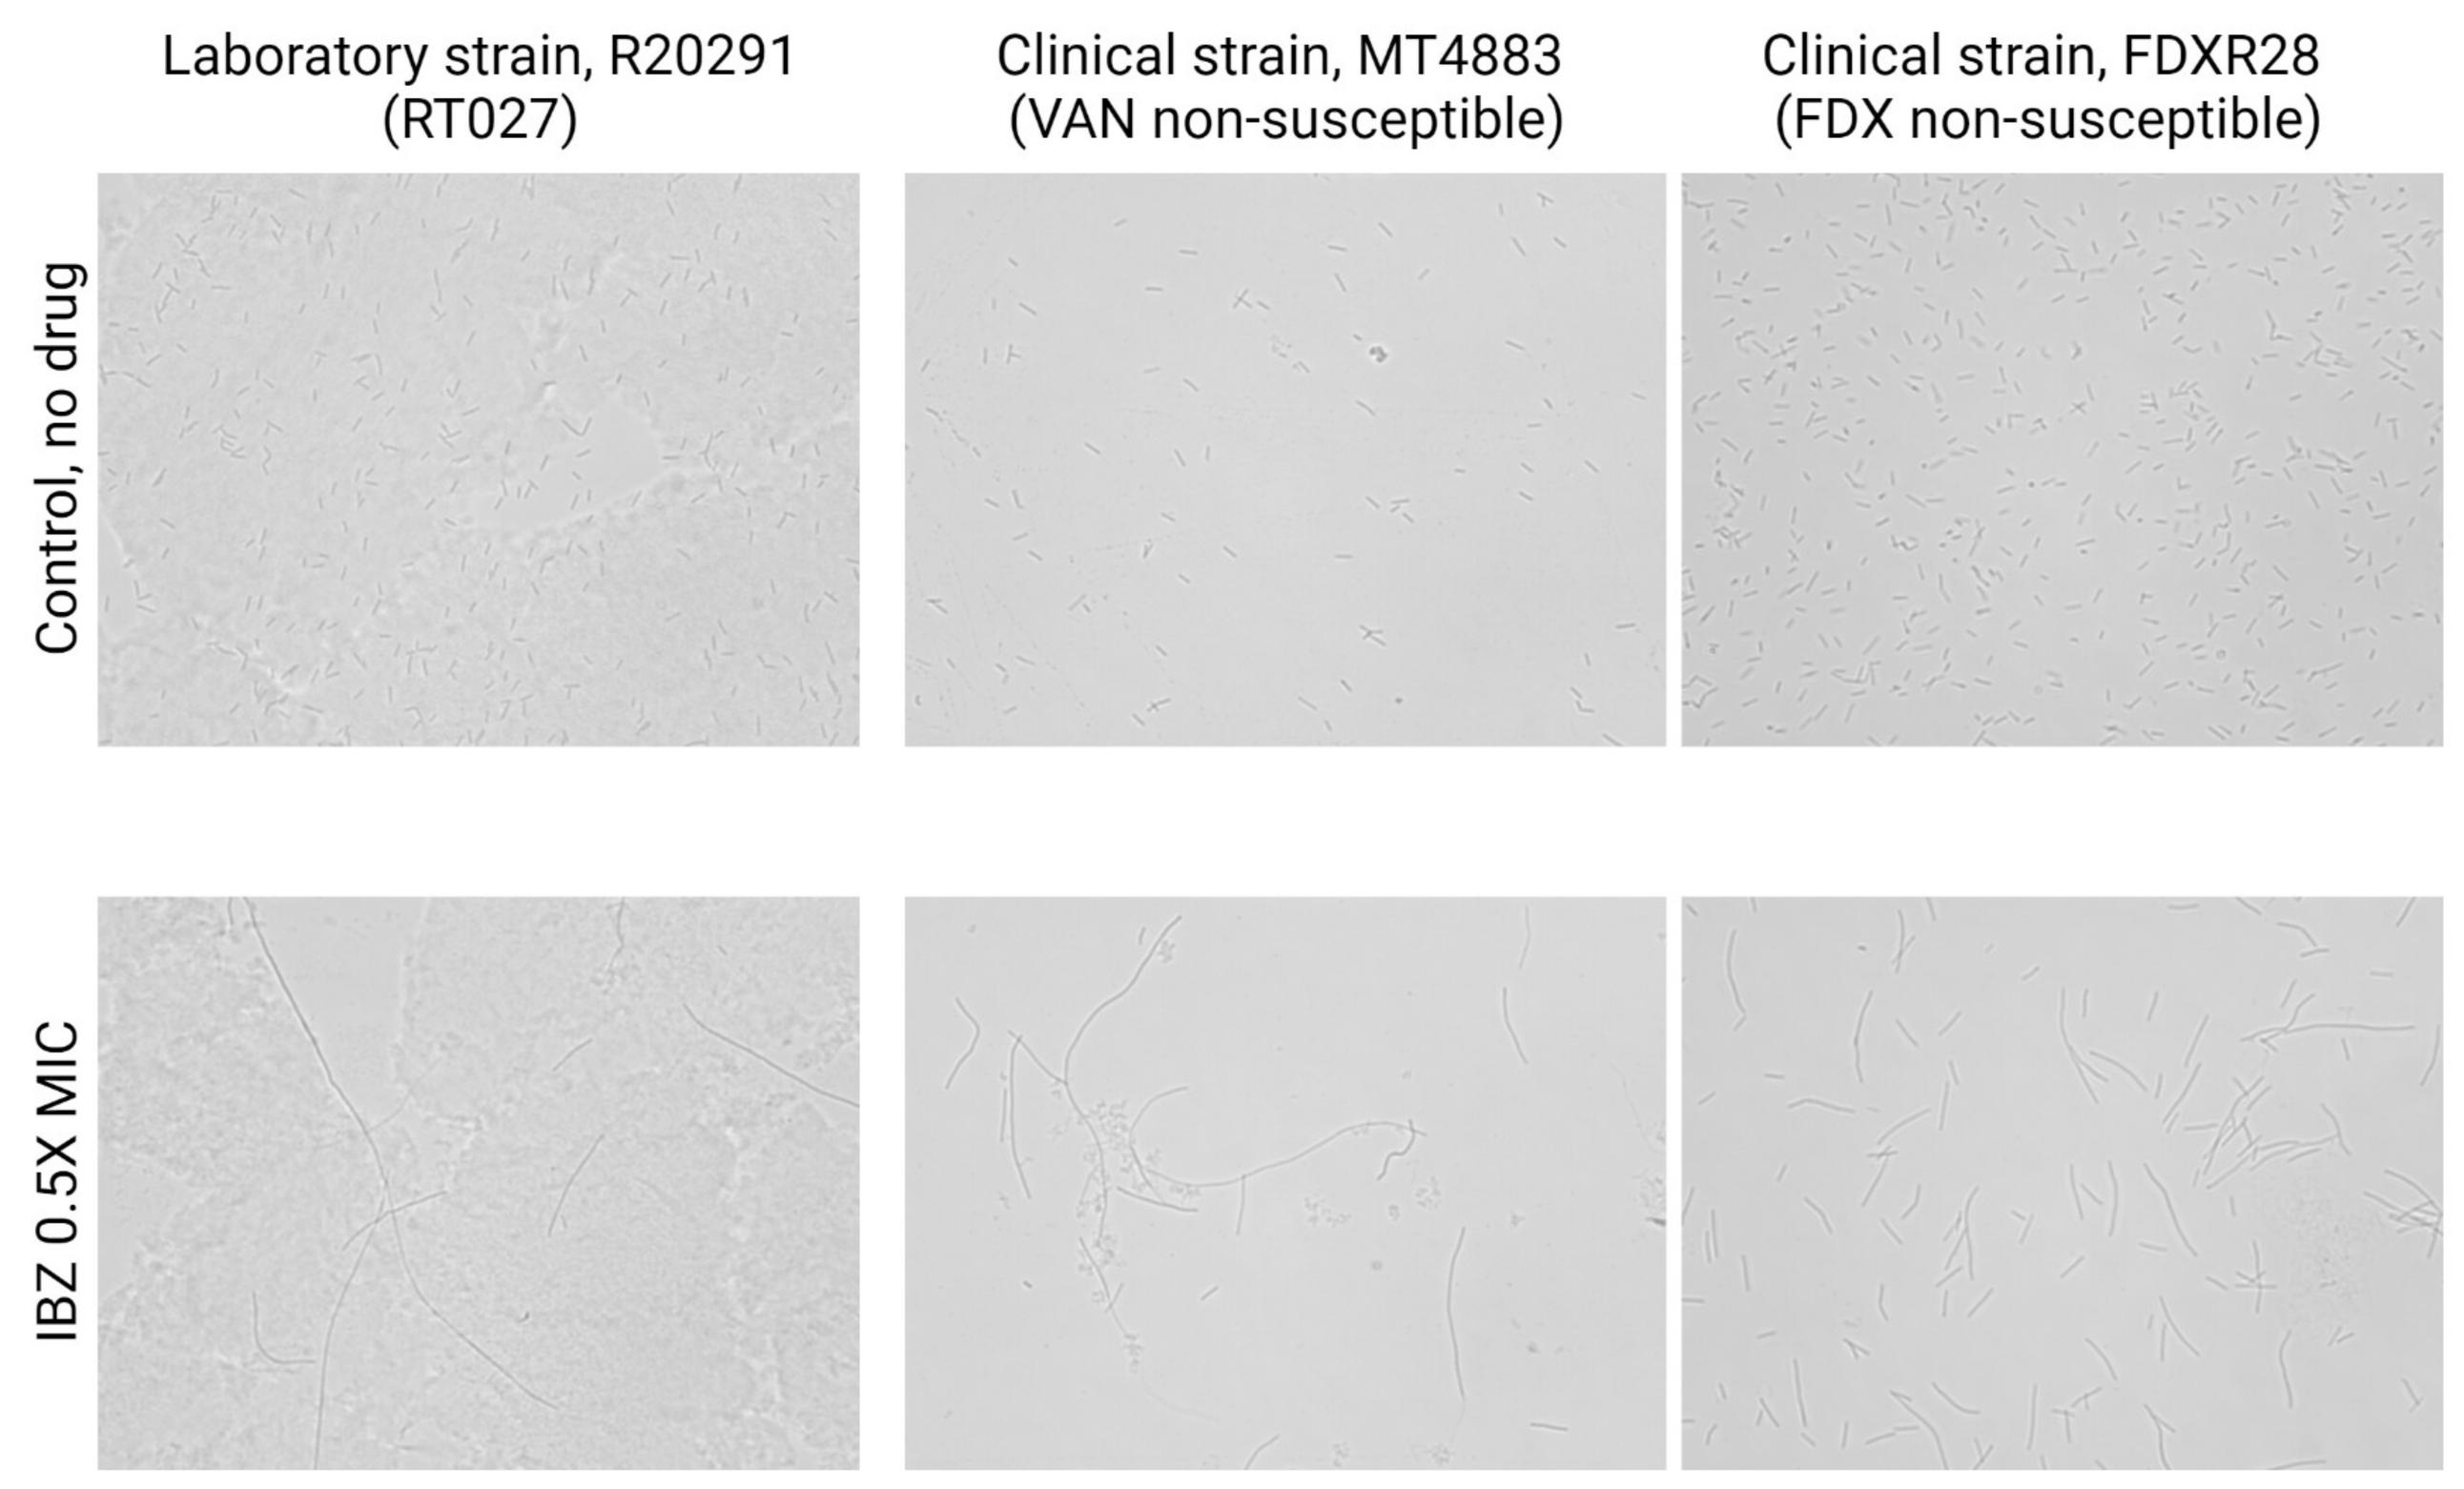
Laboratory strain, R20291 (RT027)
Clinical strain, MT4883 (VAN non-susceptible)
Clinical strain, FDXR28 (FDX non-susceptible)
Control, no drug
IBZ 0.5X MIC

CONCLUSIONS

- Ibezapolstat bactericidal activity was similar to the MIC and maintained in wild-type and non-susceptible strains.
- Time-kill assays against 2 laboratory wild-type and 2 clinical non-susceptible strains demonstrated sustained ibezapolstat activity despite reduced killing by comparator antibiotics for ibezapolstat and vancomycin non-susceptible strains.
- This study demonstrated the potent bactericidal activity of ibezapolstat against a large collection of *C. difficile* strains including multidrug-resistant strains.
- This study highlights the therapeutic potential of ibezapolstat against multidrug-resistant strains of *C. difficile*.
- The effect of ibezapolstat against multidrug-resistant strains should be further explored in animal models to take full advantage of this unique mechanism of action.

FUNDING

Investigator-initiated grant from Acurx Pharmaceuticals