Poster # 588

B-004

AACR 10/29/17

Enhanced Efficacy of Immune Modulators with Albumin Binding Domains (ABD)

Haomin Huang, Keneneshia Haenssen, Anil Bhate, Supriya Sanglikar, John Baradei, Shan Liu, Senthil Kumar, Zihao Cui, Richard Hampton, Robert Kramer and John Cini | Sonnet Biotherapeutics, 1 Duncan Drive, Cranbury, NJ 08512, USA

INTRODUCTION

Small (<100Kd) recombinant therapeutic proteins penetrate better into solid tumors but exhibit shorter circulation half-life (mins/hour vs. 21 days) when compared to IgG's. Methods to improve serum half-life for these small MW therapeutic proteins include conjugation to polyethylene glycol (ie., PEG: eg., Peg-Intron, NKT-214) or formulations on the surface of, or within, nanoparticles. Albumin has also been successfully exploited as a carrier for small therapeutic proteins. Examples include the genetic fusion of Interferon to albumin (Albuferon) and the use of a multivalent albumin binding antibody fragment that has the capacity to bind serum albumin and also neutralize TNFa (Ozoralizumab). A potential advantage to the use of albumin as a drug carrier is that albumin is known to accumulate in inflamed and angiogenic tissues such as the tumor microenvironment, and may thus provide a means of passive targeting to the tumor.

Sonnet BioTherapeutics, using a XOMA phage library, has developed a fully human single chain variable antibody (scFv) construct with demonstrated high binding affinity to serum albumin across species (mouse, human & *cynomolgus* monkey) and retains the benefits of FcRn mediated recycling of albumin for extending serum half-life. This scFv albumin binding domain (ABD) has been utilized to create several different genetic fusion constructs with various small molecular weight therapeutic proteins (e.g., recombinant interleukins such as IL12, and scFv's against relevant immune-oncology targets, ie., TGFb). In mouse models, these ABD constructs have extended serum half-lives, improved tumor accumulation and enhanced efficacy compared to their respective naked recombinant therapeutic protein. This poster summarizes the discovery of our scFv-ABD and preliminary studies with four different therapeutic scFv-ABD constructs demonstrating the broad utility of this approach.

SONNET BioTherapeutics Platform (Bi or Tri Targeting Arms)

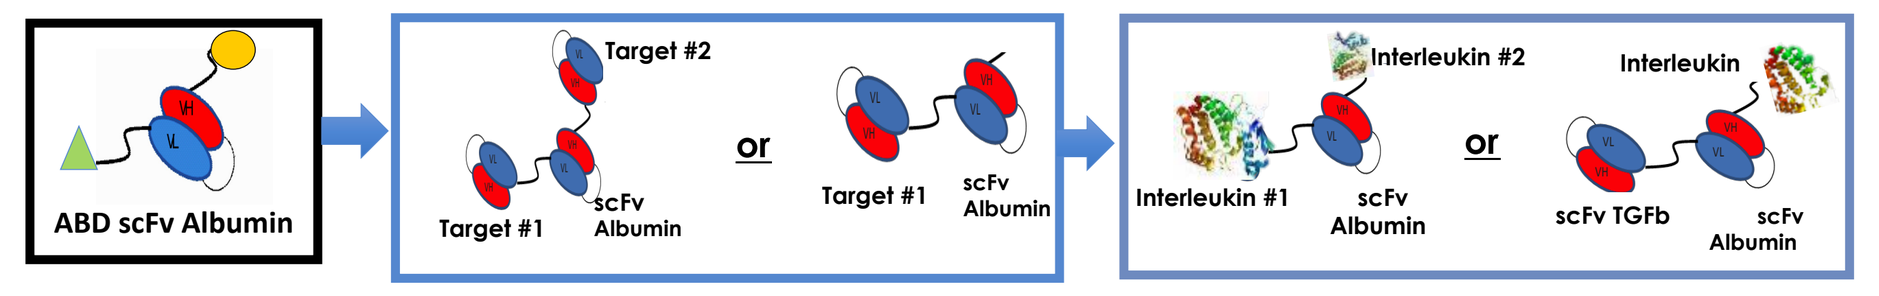
ABD scFv Albumin
Target #2
Target #1
scFv Albumin
or
Interleukin #2
Interleukin #1
Interleukin
scFv TGFb

1. **Single-Chain Antibody (ScFv) Based**
 - **Flexibility** for multiple mechanisms of action
 - **Dual affinity targets** enhance specific tumor delivery
 - **Small size** enhanced tumor penetration
 - **Plug-and-play versatility** for target selection can *inhibit and/or stimulate*
2. **Albumin Binding Domain (ABD); scFv binds Human Serum Albumin (HSA)**
 - **Tumor delivery –** HSA is elevated in tumors & inflamed tissues
 - **Enhanced pharmacokinetics -** $T^{1/2}$ from minutes to days
3. **Recombinant Human (Hu) Proteins (Interleukins; scFv vs. TGFb)**
 - ABD – hu Interleukins (e.g. IL12 or IL15)
 - ABD – 2X hu Interleukins (e.g. IL15 –ABD- IL12)
 - ABD – scFv (e.g. TGF β)

Figure 1

ABD Bio-Activity Selection Criteria to Ensure Increase in PK

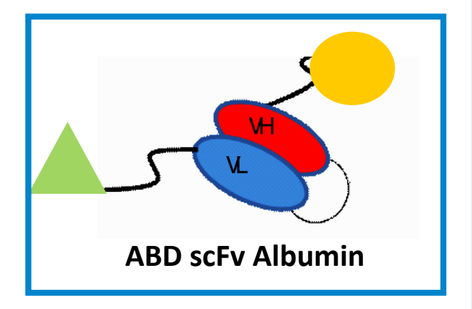
ABD scFv Albumin

- ✔ Binding to Human & Cyno Serum Albumin *kD ~20 - 60 nM*
- ✔ Binding to Mouse Serum Albumin *kD ~10 - 30 nM*
- ✔ Binding at low pH 5.5 & neutral pH7.2 for human, mouse albumin
- ✔ No competition with FcRn binding to serum albumin. In-vitro studies with ABD showed cell recycling with the FcRn hence should extend pK in-vivo (see Fig below)
- ✔ Binds Albumin in serum by immune-precipitation
- ✔ Human serum stability at 37C for 7-14 Days
- ✔ Immunogenicity tested by Antitope computer analysis

XOMA (Library 47) full human scFv phage library was used to screen for a scFv by solid & solution panning against HSA, MSA and h/mTGFβ 1, 2 & 3.

ABD selection was based on meeting all of the above criteria.

Anti Albumin Binding Domain (ABD) A10M3 is a scFv: VH3/VL1, Tm ~60C, MW 26KD, pI 7.1

Anti TGFβ selection based on blocking TGF inducing of: Tregs proliferation, TGF→SMAD2 PO4, EMT & Migration

Anti TGFβ (4D9M) is a scFv: VH3/VL1, Tm ~62C, MW 26KD, pI 5.3

Figure 2

ABD Binding to Albumin/FcRn in Live Cells Escape Lysosomal Degradation

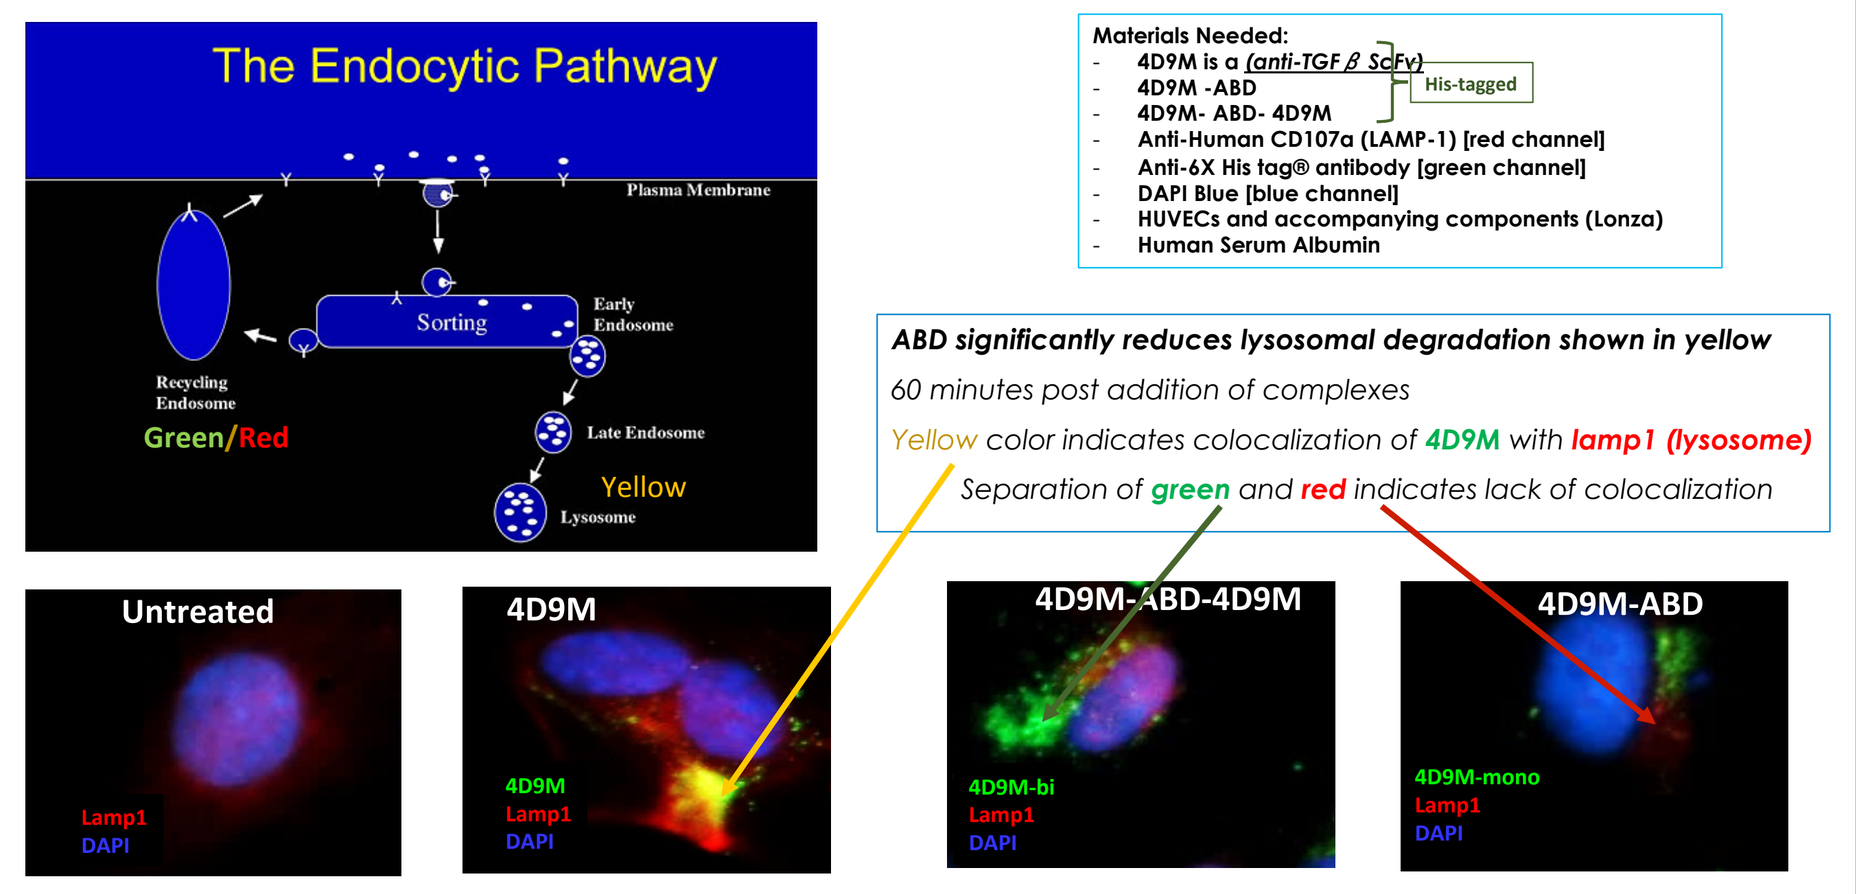
The Endocytic Pathway
Plasma Membrane
Sorting
Early Endosome
Recycling Endosome
Green/Red
Late Endosome
Yellow
Lysosome
Materials Needed:
4D9M is a (anti-TGF β ScFv)
4D9M -ABD
4D9M- ABD- 4D9M
His-tagged
Anti-Human CD107a (LAMP-1) [red channel]
Anti-6X His tag® antibody [green channel]
DAPI Blue [blue channel]
HUVECs and accompanying components (Lonza)
Human Serum Albumin
ABD significantly reduces lysosomal degradation shown in yellow
60 minutes post addition of complexes
Yellow color indicates colocalization of 4D9M with lamp1 (lysosome)
Separation of green and red indicates lack of colocalization
Untreated
4D9M
4D9M-ABD-4D9M
4D9M-ABD
Lamp1
DAPI
4D9M-bi
4D9M-mono

Figure 3

Evaluation of anti-TGF β 4D9M scFv & anti-TGF β 4D9M-ABD Tumor Accumulation

In vivo Proof of Concept

Western blot analysis of Mouse 4T1 (TGF β positive) tumor (@ 100 mm³) extracts from mice terminated at 0.5, 4, 12 and 24 hours post IV injection with 100 ug/mouse of: ABD, antiTGF β or antiTGF β -ABD.

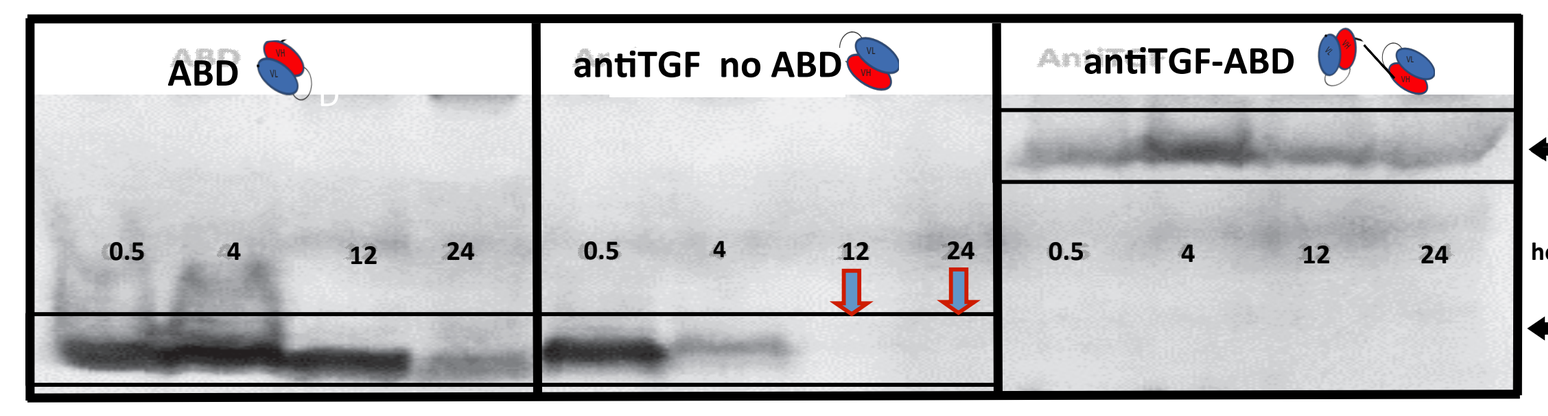
ABD
antiTGF no ABD
antiTGF-ABD
0.5
4
12
24
50kD
hours
25 kD

Results show accumulation and retention of ABD in the tumor
- ABD - Present at 0.5 hours, peaks at 4 hours and detectable through 24 hours.
- Anti-TGF svFv - Present at 0.5 hours then declines at 4 hours and undetectable at 12 and 24 hours.
- Anti-TGF svFv-ABD – is present at 0.5 hours. and detectable through 24 hours.

ABD accumulates in the tumors 24 hrs+ and without ABD, the scFv enters tumor but diffuses out after 4 hrs.

Data supports ABD POC for the platform's ability to enhance penetration/accumulation & retention within the tumor.

Figure 4

PK Half-Life $T^{1/2}$ of ABD Constructs IL12 & IL15

The aim of this study is to demonstrate in mice, the pharmacokinetic (PK) behavior of naked IL12 & IL15 compared to the same fusion proteins linked to albumin binding domain (ABD) IL12-ABD & IL15-ABD.

Method: 8 mice C57B/ TP, Age 9.5 weeks dose IV, sacrificed @ 5, 15, 30 mins, 1, 2, 4, 8, 24 & 48 hrs. Serum tested by ELISA

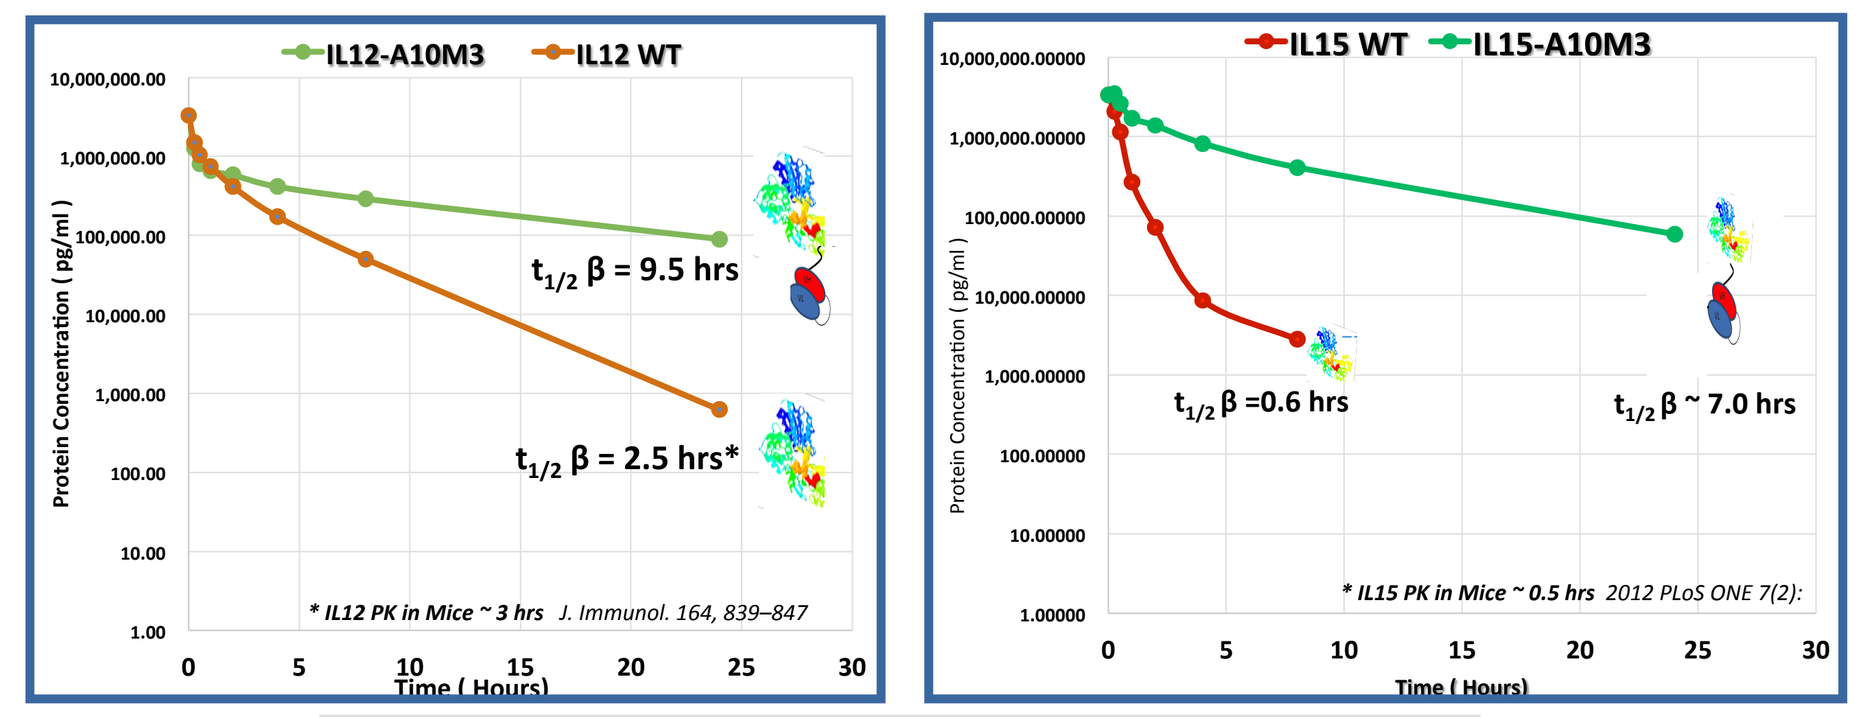
IL12-A10M3
IL12 WT
$t_{1/2}$ β = 9.5 hrs
$t_{1/2}$ β = 2.5 hrs*
* IL12 PK in Mice ~ 3 hrs J. Immunol. 164, 839–847
IL15 WT
IL15-A10M3
$t_{1/2}$ β =0.6 hrs
$t_{1/2}$ β ~ 7.0 hrs
* IL15 PK in Mice ~ 0.5 hrs 2012 PLoS ONE 7(2):
Protein Concentration (pg/ml)
Time (Hours)

Fusion to ABD increased the plasma half-life of IL12 > 4x and IL15 >10X
IL12 MW = 70 kd vs IL15 MW=13 kd

Figure 5

Effect of IL-15 ABD in B16F10 on Tumor Growth and on Lymphocyte Populations in Spleens and Tumors

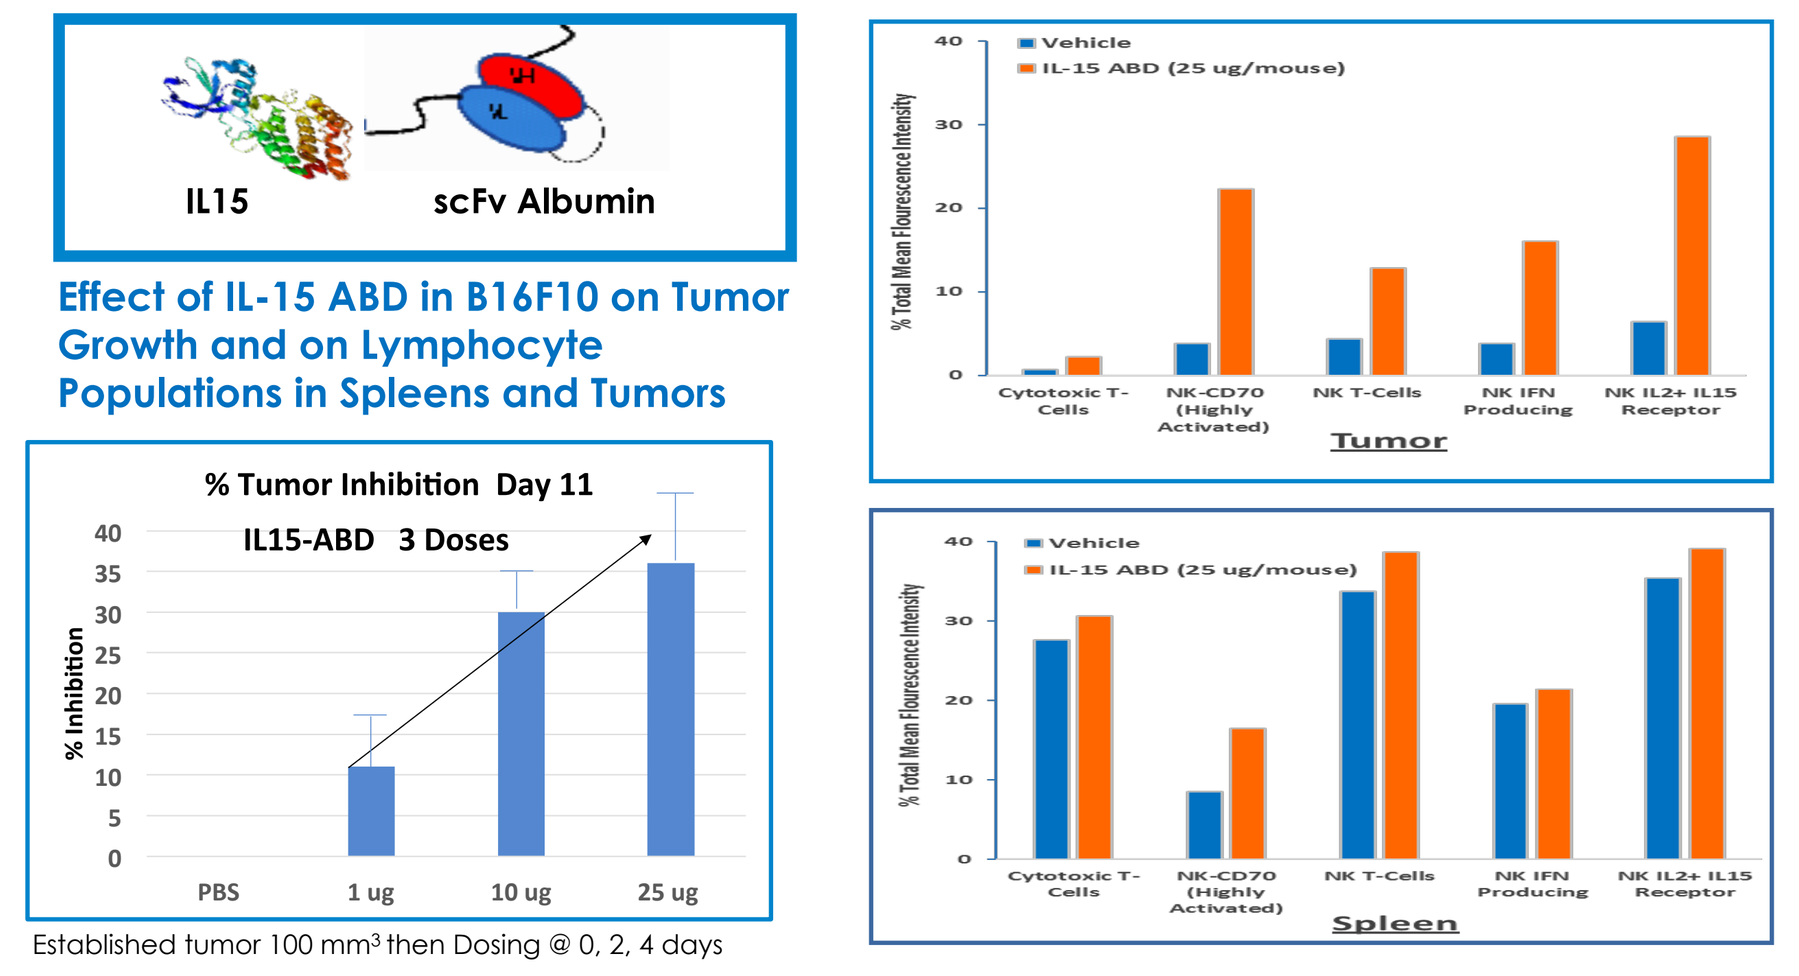
IL15
scFv Albumin
% Tumor Inhibition Day 11
IL15-ABD 3 Doses
% Inhibition
PBS
1 ug
10 ug
25 ug
Vehicle
IL-15 ABD (25 ug/mouse)
% Total Mean Flourescence Intensity
Cytotoxic T-Cells
NK-CD70 (Highly Activated)
NK T-Cells
NK IFN Producing
NK IL2+ IL15 Receptor
Tumor
Spleen

Established tumor 100 mm³ then Dosing @ 0, 2, 4 days

IL15-ABD showed reduction in TV and a 5-7 fold increase in the tumor NK cells (FACS data next slide)

Figure 6

Effect of IL-15 ABD Treatment on Tumor Infiltration Lymphocyte Populations

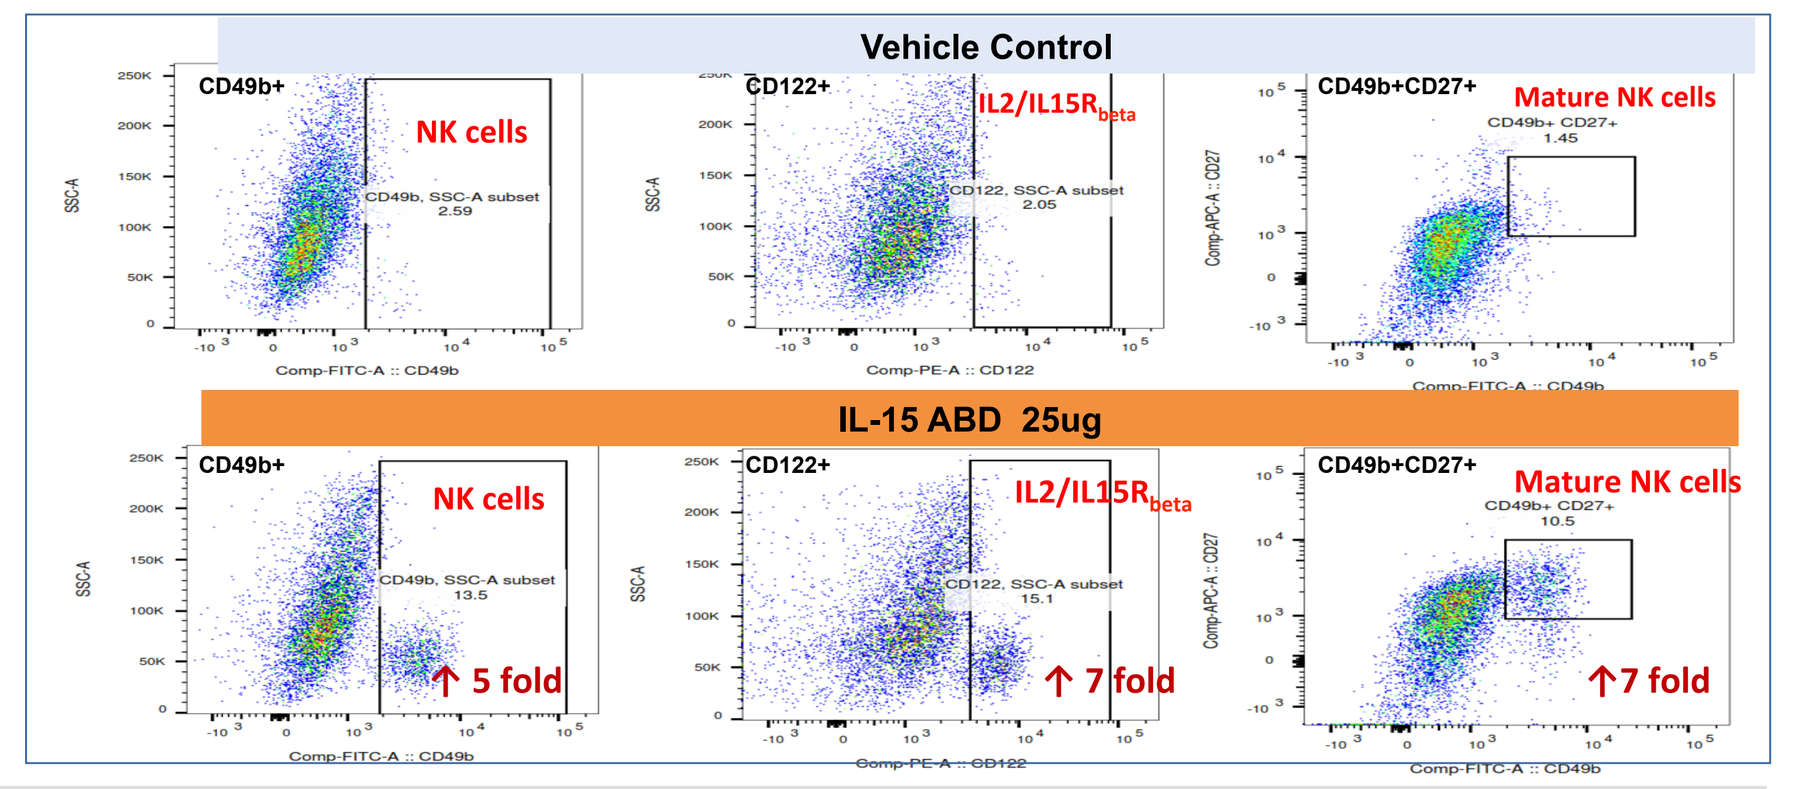
Vehicle Control
IL-15 ABD 25ug
CD49b+
CD122+
CD49b+CD27+
NK cells
IL2/IL15Rbeta
Mature NK cells
↑ 5 fold
↑ 7 fold
↑7 fold

Analysis of infiltrating lymphocyte populations by FACS show increases in NK cell populations within the tumor of IL15-ABD treated mice resulting in a reduction of tumor growth.

Figure 7

Day 10 Post Single Dose (@ 100 mm³) of IL-12 or IL-12-ABD in B16F10 Melanoma

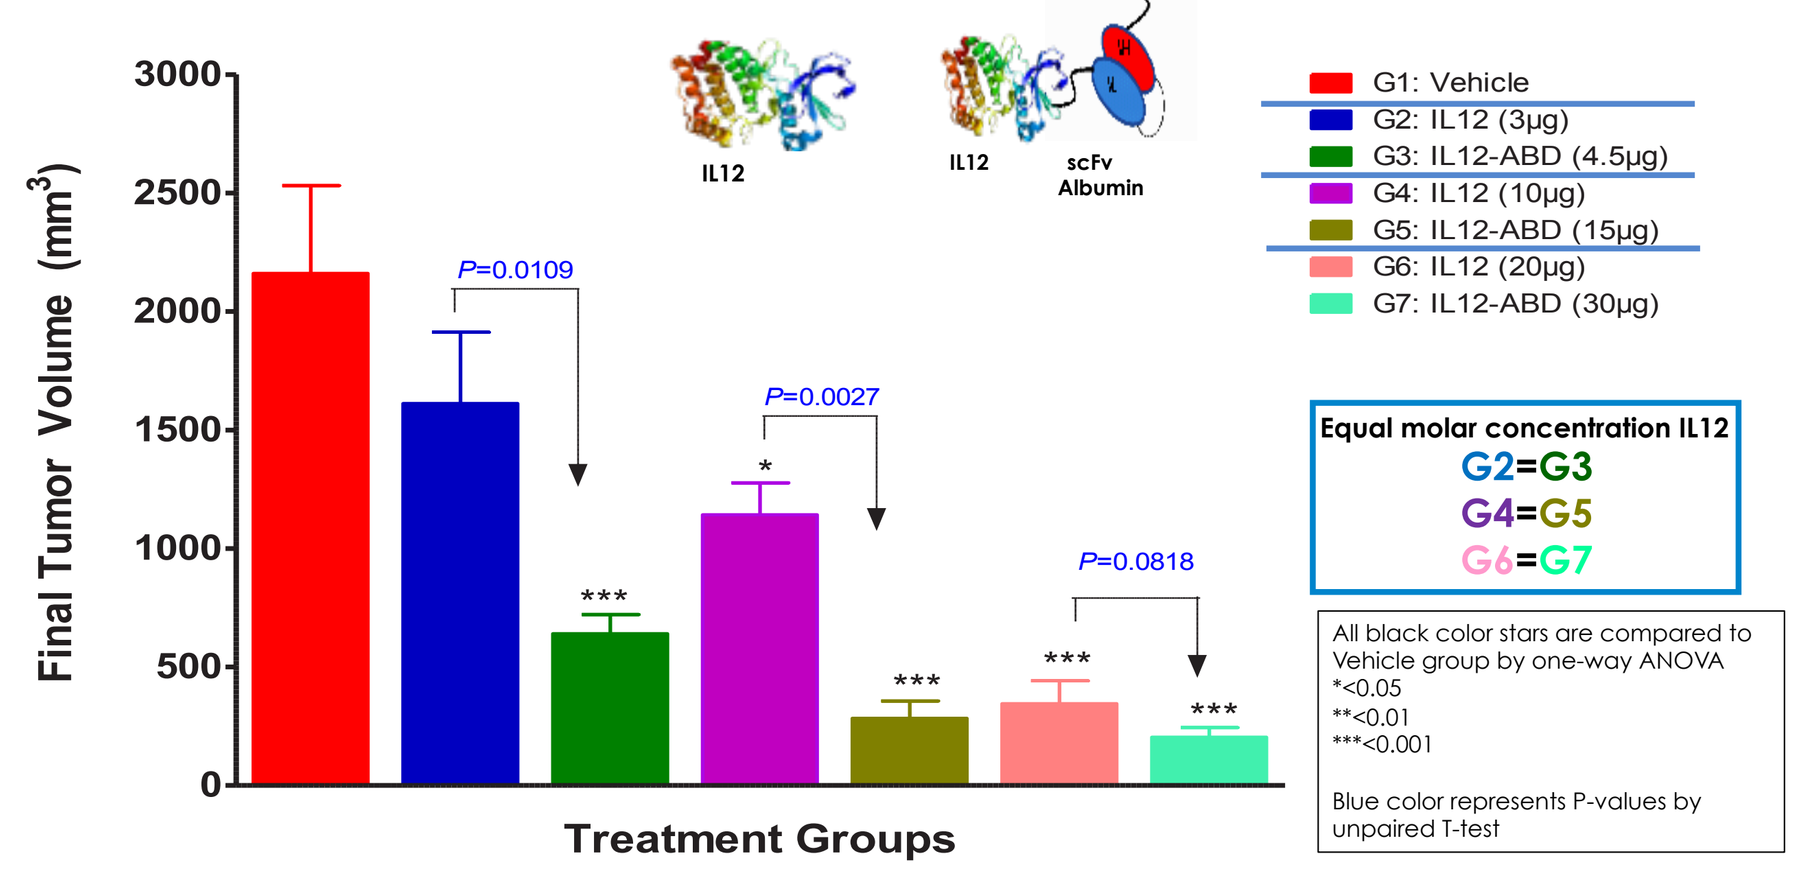
Final Tumor Volume (mm³)
Treatment Groups
IL12
scFv Albumin
G1: Vehicle
G2: IL12 (3μg)
G3: IL12-ABD (4.5μg)
G4: IL12 (10μg)
G5: IL12-ABD (15μg)
G6: IL12 (20μg)
G7: IL12-ABD (30μg)
P=0.0109
P=0.0027
P=0.0818
Equal molar concentration IL12
G2=G3
G4=G5
G6=G7
All black color stars are compared to Vehicle group by one-way ANOVA
*<0.05
**<0.01
***<0.001
Blue color represents P-values by unpaired T-test

Equal molar concentration of IL12 show that IL12-ABD is more potent than IL12

Figure 8

Comparison of Pharmacodynamic Effects of IL12-ABD & IL12 @ 5 Days After Single Dose B16-F10 Tumor-bearing (100 mm³)

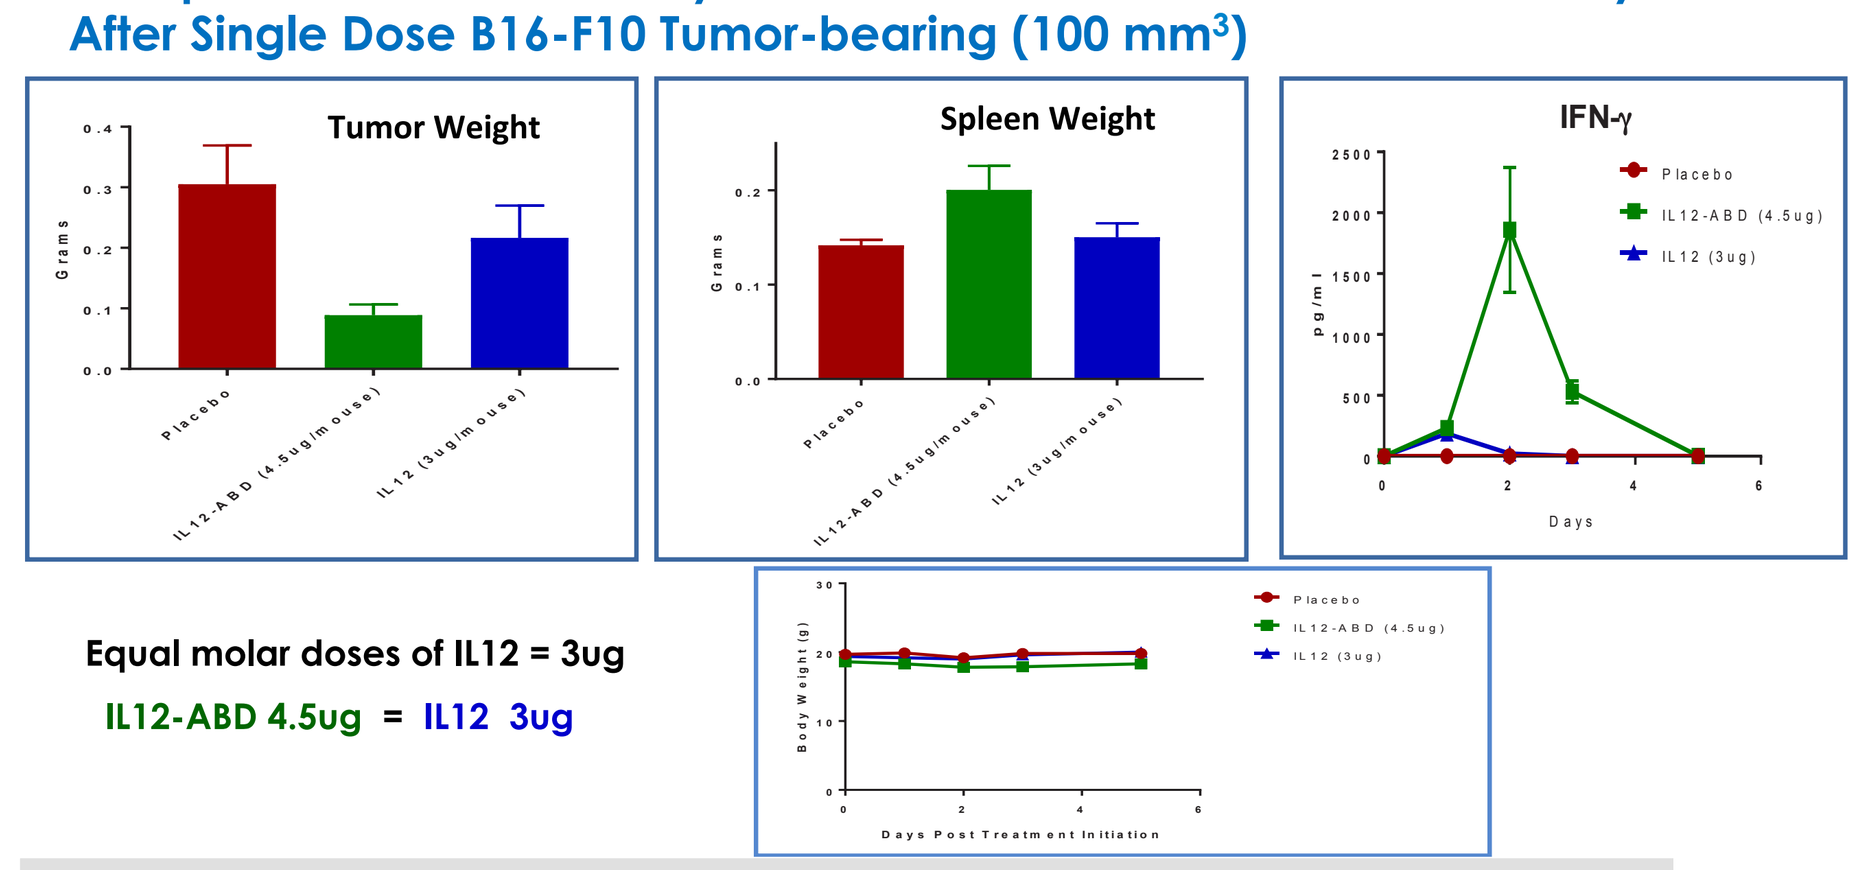
Tumor Weight
Spleen Weight
IFN-γ
Placebo
IL12-ABD (4.5ug)
IL12 (3ug)
Days
Equal molar doses of IL12 = 3ug
IL12-ABD 4.5ug = IL12 3ug

IL12-ABD demonstrated a greater suppression of tumor growth and a corresponding increase in immune activation as shown by the increase in spleen weight and a transient increase in serum IFN without an effect on mouse bodyweight compared to the molar equivalent dose of recombinant IL12.

Figure 9

Evaluation of Single Dose (SD) IL12-ABD (1.3 ug) vs. IL12 (30ug) in B16F10 Melanoma (established @100 mm³ , n=8)

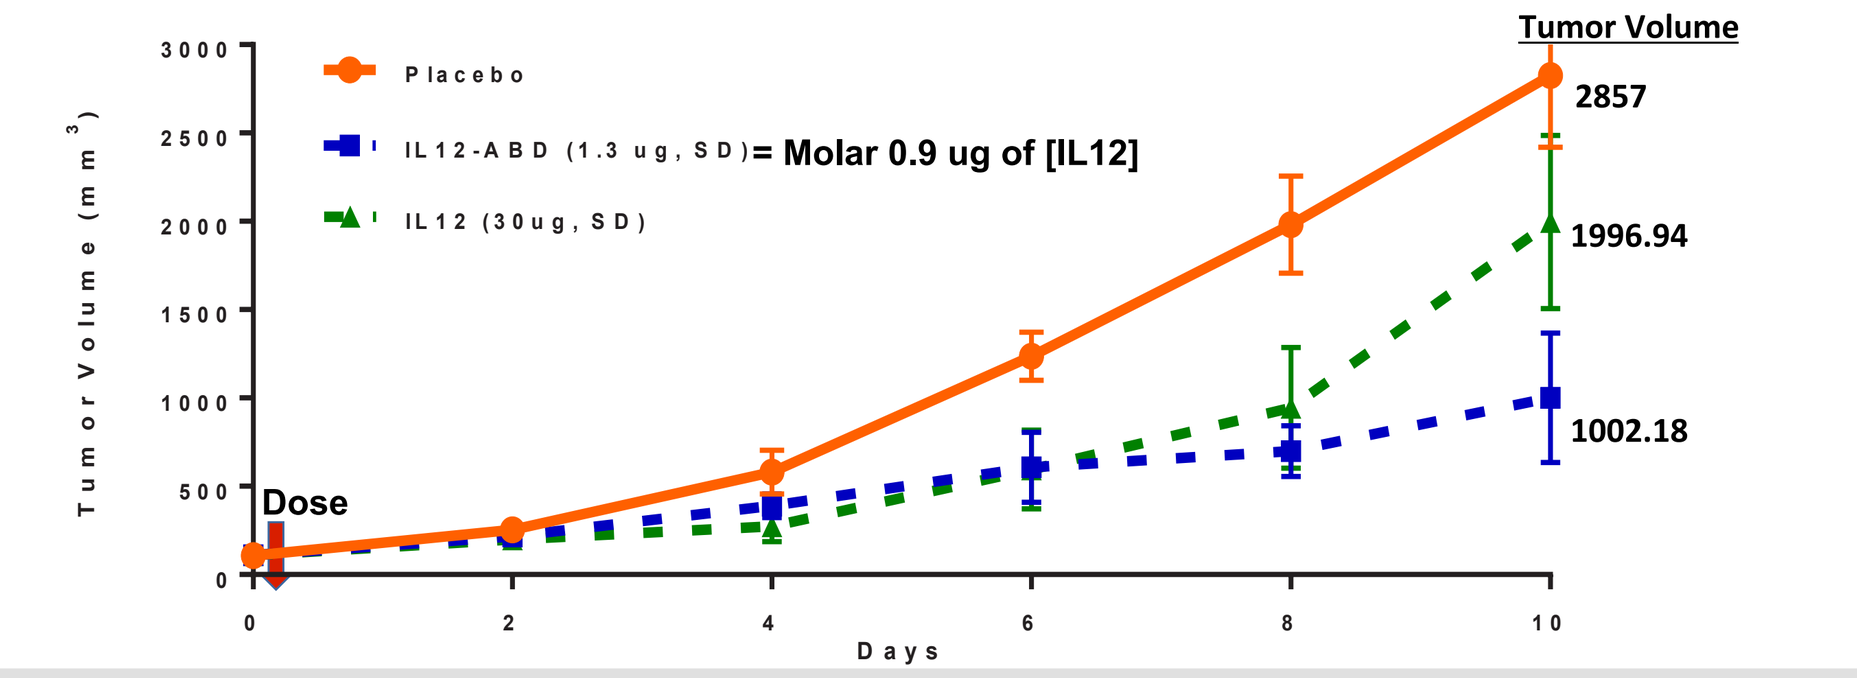
Tumor Volume
Placebo
IL12-ABD (1.3 ug, SD) = Molar 0.9 ug of [IL12]
IL12 (30ug, SD)
2857
1996.94
1002.18
Dose
Days
Tumor Volume (mm³)

Recombinant IL12 (1ug) & IL12-ABD (1.3ug) are molar equivalent & have the same bioactivity in-vitro (data not shown). However, IL12-ABD was ~ 30+ fold more potent than recombinant IL12 in vivo as reflected by a single dose of IL12-ABD at 1.3 ug producing a comparable reduction in tumor volume as a single 30 ug dose of recombinant IL12 on days 1-8 post treatment. The duration of benefit was longer for IL12-ABD (day 10). Hematologic changes and serum IFN γ are reported in Figure 11.

Figure 10

Comparison of Hematopoietic Effects of IL12-ABD (1.3ug) & IL12 (30ug) at 3 & 7 Days After Single Dose B16-F10 Tumor-bearing (100 mm³)

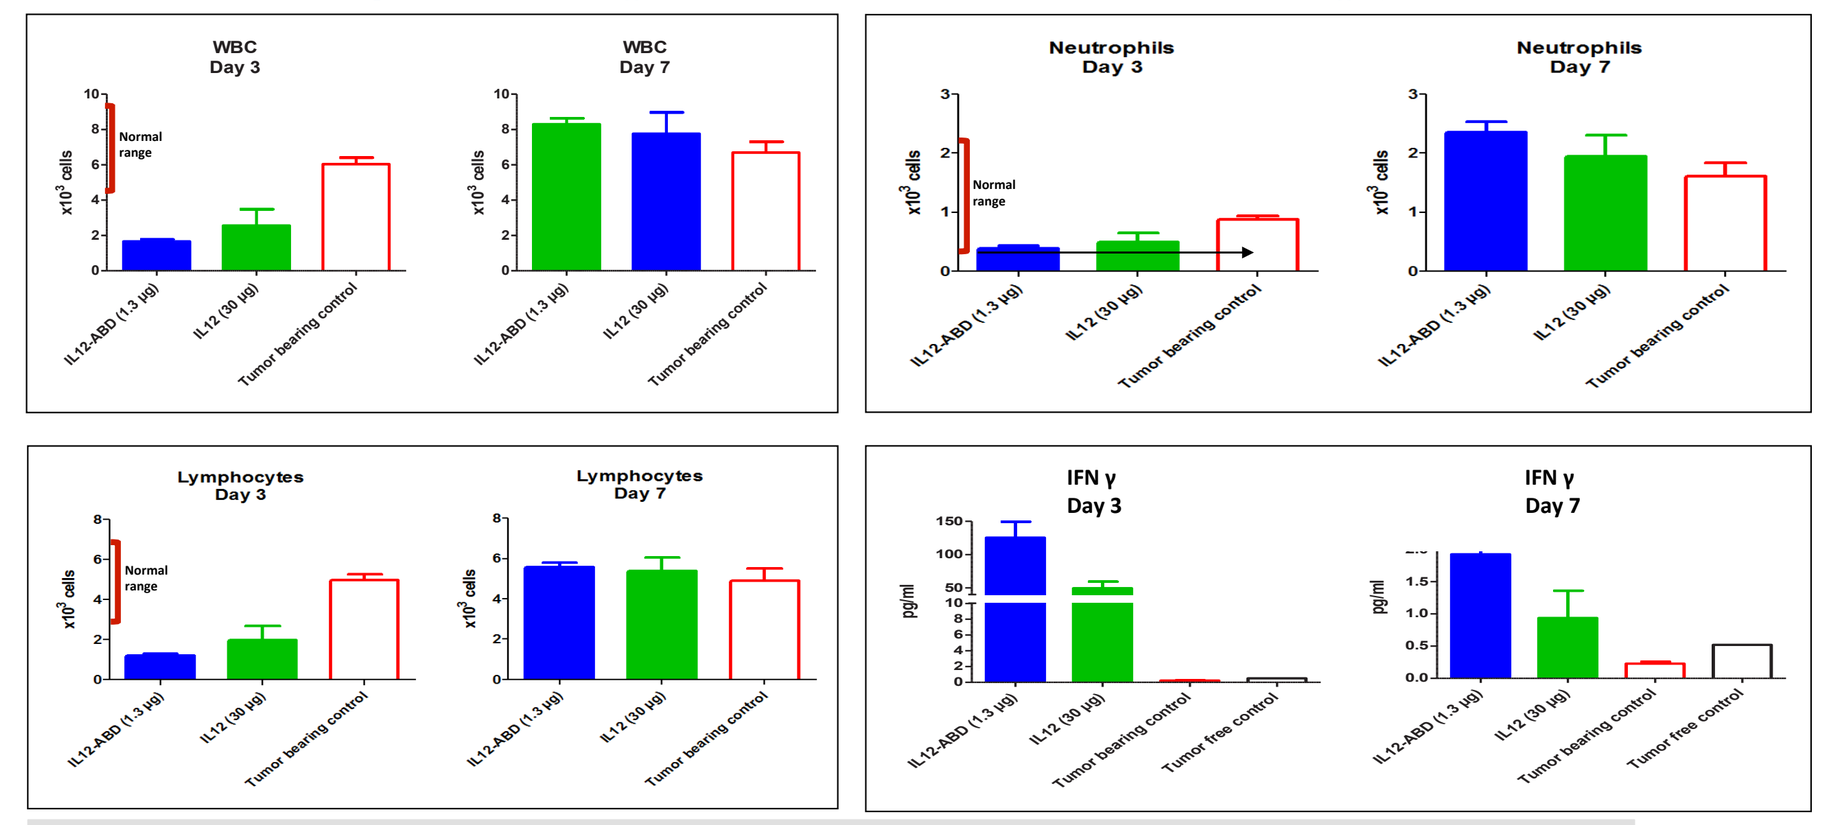
WBC Day 3
WBC Day 7
Neutrophils Day 3
Neutrophils Day 7
Lymphocytes Day 3
Lymphocytes Day 7
IFN γ Day 3
IFN γ Day 7
Normal range
IL12-ABD (1.3 μg)
IL12 (30 μg)
Tumor bearing control
Tumor free control

IL12-ABD (1.3 ug) and recombinant IL12 (30ug) produced comparable transient reductions in hematologic parameters that returned to control values by day 7. However, IL12-ABD produced a significantly greater transient increase in serum IFN γ on day 3, that was comparable to recombinant IL12 IFN γserum values by day 7.

Figure 11

Sonnet Tri-Specific Construct

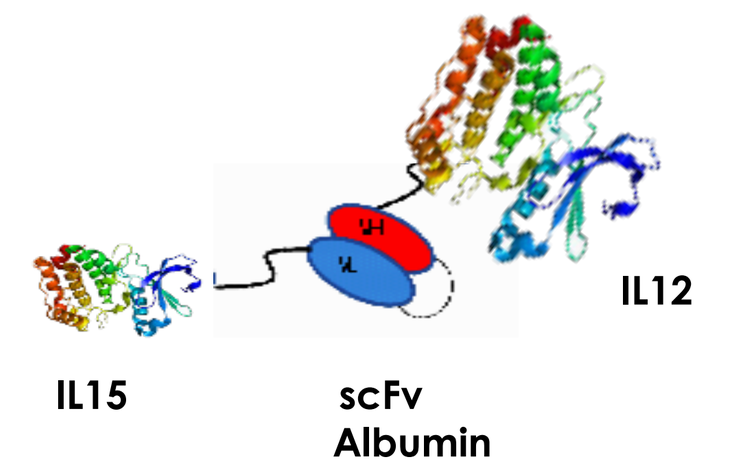
IL15
scFv Albumin
IL12

Enhanced Biologic Activity:

IL12 : ↑ IL15 alpha receptor, IFN, NK/Tcells, TH1 & ↓ Treg

IL15 : ↑ IL12 beta 1 receptor, NK cells & ↓ CD8 memory loss

Figure 12

Assessment of IL-12 & IL15 Cell Based Activity for the IL15-ABD-IL12

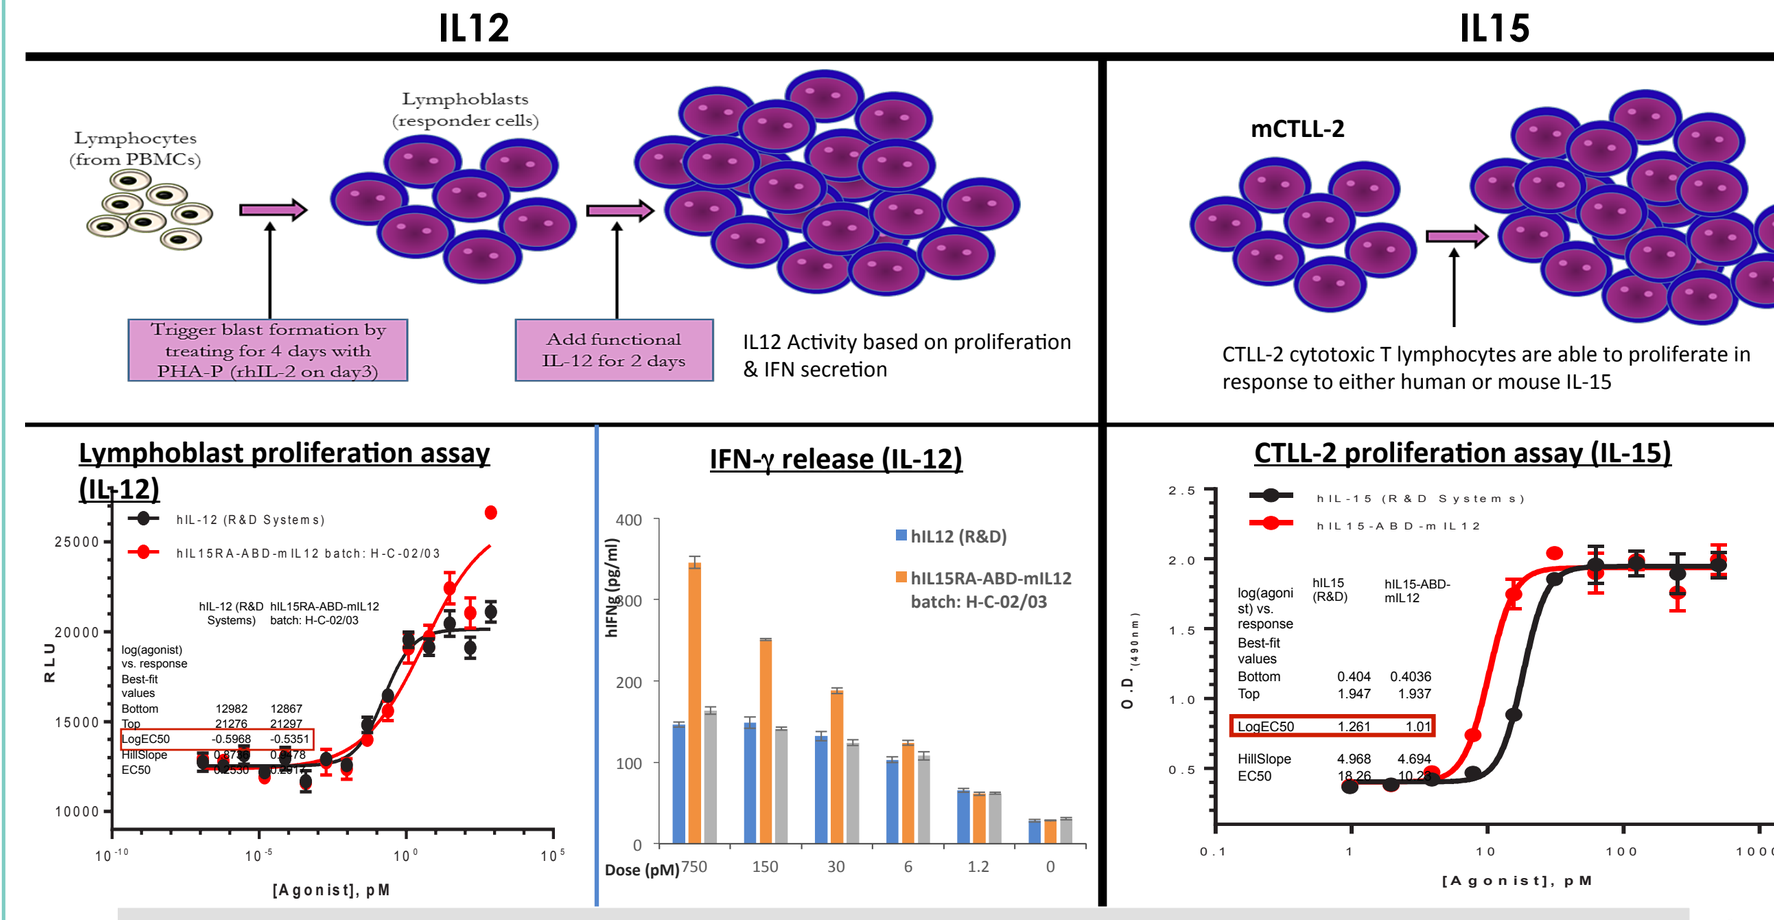
IL12
IL15
Lymphocytes (from PBMCs)
Lymphoblasts (responder cells)
Trigger blast formation by treating for 4 days with PHA-P (rhIL-2 on day3)
Add functional IL-12 for 2 days
IL12 Activity based on proliferation & IFN secretion
mCTLL-2
CTLL-2 cytotoxic T lymphocytes are able to proliferate in response to either human or mouse IL-15
Lymphoblast proliferation assay (IL-12)
IFN-γ release (IL-12)
CTLL-2 proliferation assay (IL-15)
[Agonist], pM

IL15-ABD-IL12 cell based assays show bio-activity in the tri-specific construct

Figure 13

IL12-ABD vs. IL15-ABD-IL12 @ Equal Molar Concentration to the IL12 Single Dose (@ 100 mm³) Day 10 Tumor Volume (n=8)

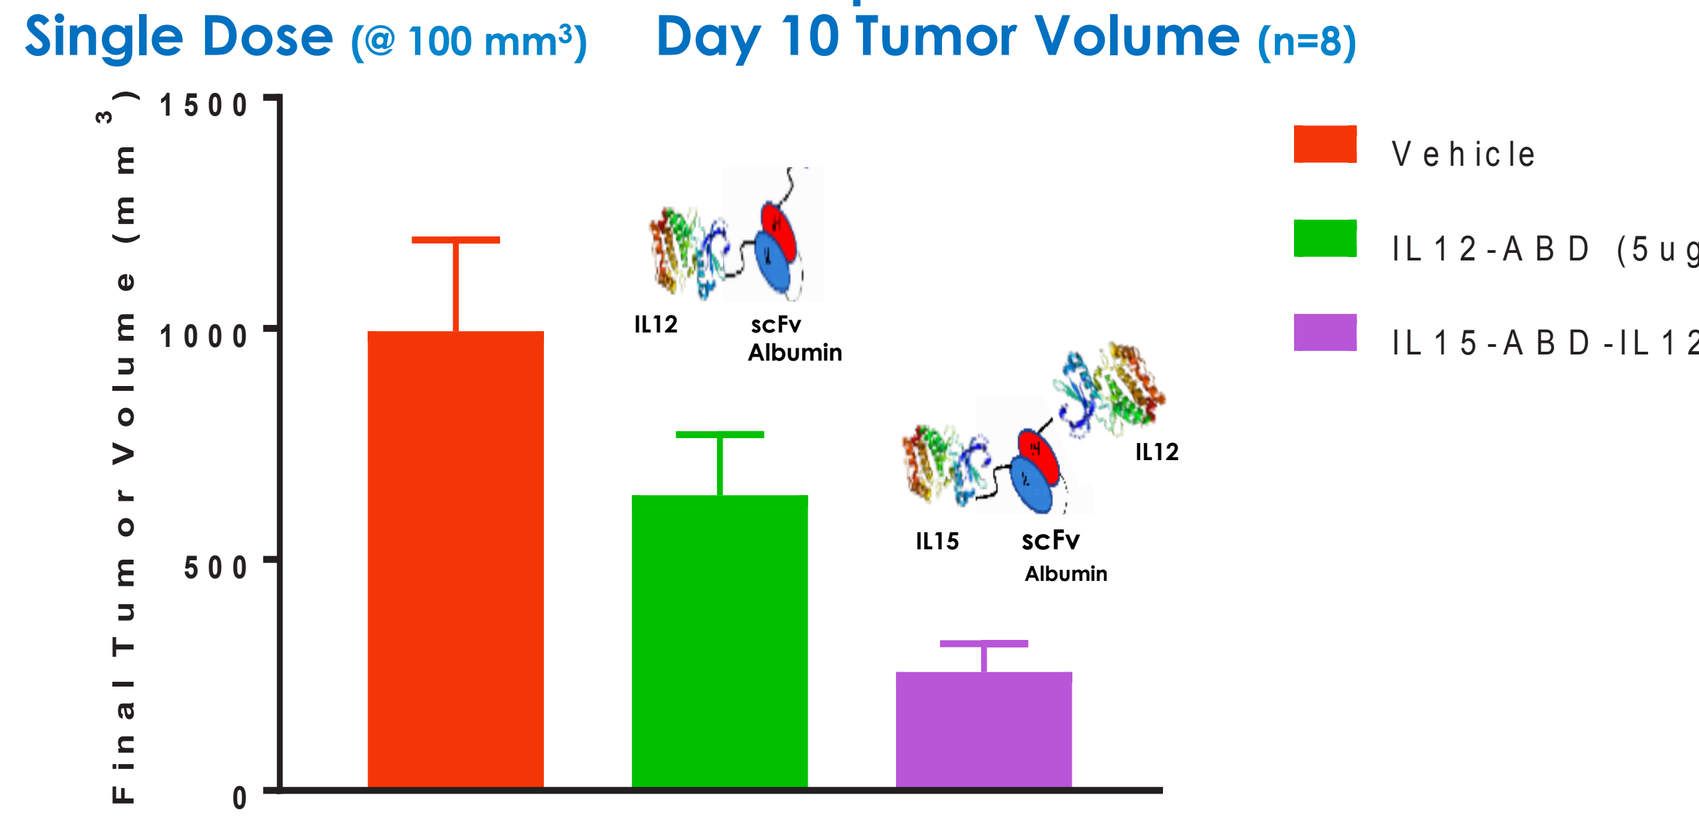
Final Tumor Volume (mm³)
Vehicle
IL12-ABD (5ug)
IL15-ABD-IL12 (6ug)
IL12
scFv Albumin
IL15

The molar equivalent dose of IL12 used in the IL12-ABD-IL15 (6ug) construct was more effective than IL12-ABD (5ug) vs the established B16F10 sc mouse melanoma tumor model. Other in-vivo studies show that free IL12 (5ug) + IL15 (1ug) is less than 50% as effective as IL15-ABD-IL12 (6ug)(not shown).

Figure 14

CONCLUSIONS

1. A novel 26 Kd scFv antibody (ABD) that binds mouse, *cynomolgus* monkey and human serum albumin has been identified, and is being developed, as a platform for enhancing the pharmacokinetics and tumor targeting of small molecular weight therapeutic proteins (Figures 1-3).
2. We demonstrate increased retention of an anti-TGFβ-scFv-ABD construct in 4T1 tumors in mice (Figure 4) and up to a 10-fold enhancement of the serum half-life in mice of recombinant IL12 or IL15 when fused to the scFv-ABD construct (Figure 5).
3. Dose dependent increases in anti-tumor activity were demonstrated with either IL15-ABD or IL12-ABD constructs (Figures 6-8), producing a corresponding increase in tumor infiltrating NK cells for IL15-ABD (Figures 6 & 7). FACs studies on cell infiltrates following IL12-ABD treatment are planned.
4. Molar equivalent doses of IL12-ABD were significantly more efficacious (3-30 fold) than naked recombinant IL12 vs. the B16F10 mouse melanoma model (Figure 8 & 10) and produced a corresponding increase in immune response as reflected by spleen weight and serum IFN, which was transient and had no effect on mouse body weight (Figure 9).
5. A first-in-class dual targeting tri-specific cytokine construct (IL15-ABD-IL12) demonstrated slightly enhanced in vitro cellular activity compared to either recombinant IL12 or IL15 (Figure 13), that translated to superior in vivo activity compared to IL12-ABD vs. the B16F10 mouse melanoma model (Figure 14).

Sonnet wishes to acknowledge Drs. Olesia Buiakova & Yan Xu (InVivoTek, NJ) for their excellent work involving all animal studies,

www.sonnetbio.com

info@sonnetbio.com