Enhancing NK cell function in the 'cold' tumor microenvironment of prostate cancer with a novel Tri-specific Killer Engager against prostate-specific membrane antigen (PSMA)

Shee Kwan (Gwen) Phung[1], Yvette Soignier[1], Nicholas Zorko[1], Trygve Nelson[1], Joshua Walker[1], Philippa R. Kennedy[1], Jeffrey S. Miller[1], Martin Felices[1]

[1]University of Minnesota, Department of Medicine, Minneapolis, MN, 55455, USA

Introduction

Natural killer (NK) cell effector function is suppressed in the tumor microenvironment (TME) of metastatic castration-resistant prostate cancer (mCRPC), the lethal form of prostate cancer. This is largely due to the inherently 'cold' nature of the TME of mCRPC that is hypoxic and has limited infiltration of cytolytic lymphocyte. In addition, immuno-suppressive cells such as myeloid-derived suppressor cells (MDSC) found in the TME also play a role in impairing NK cell effector function.

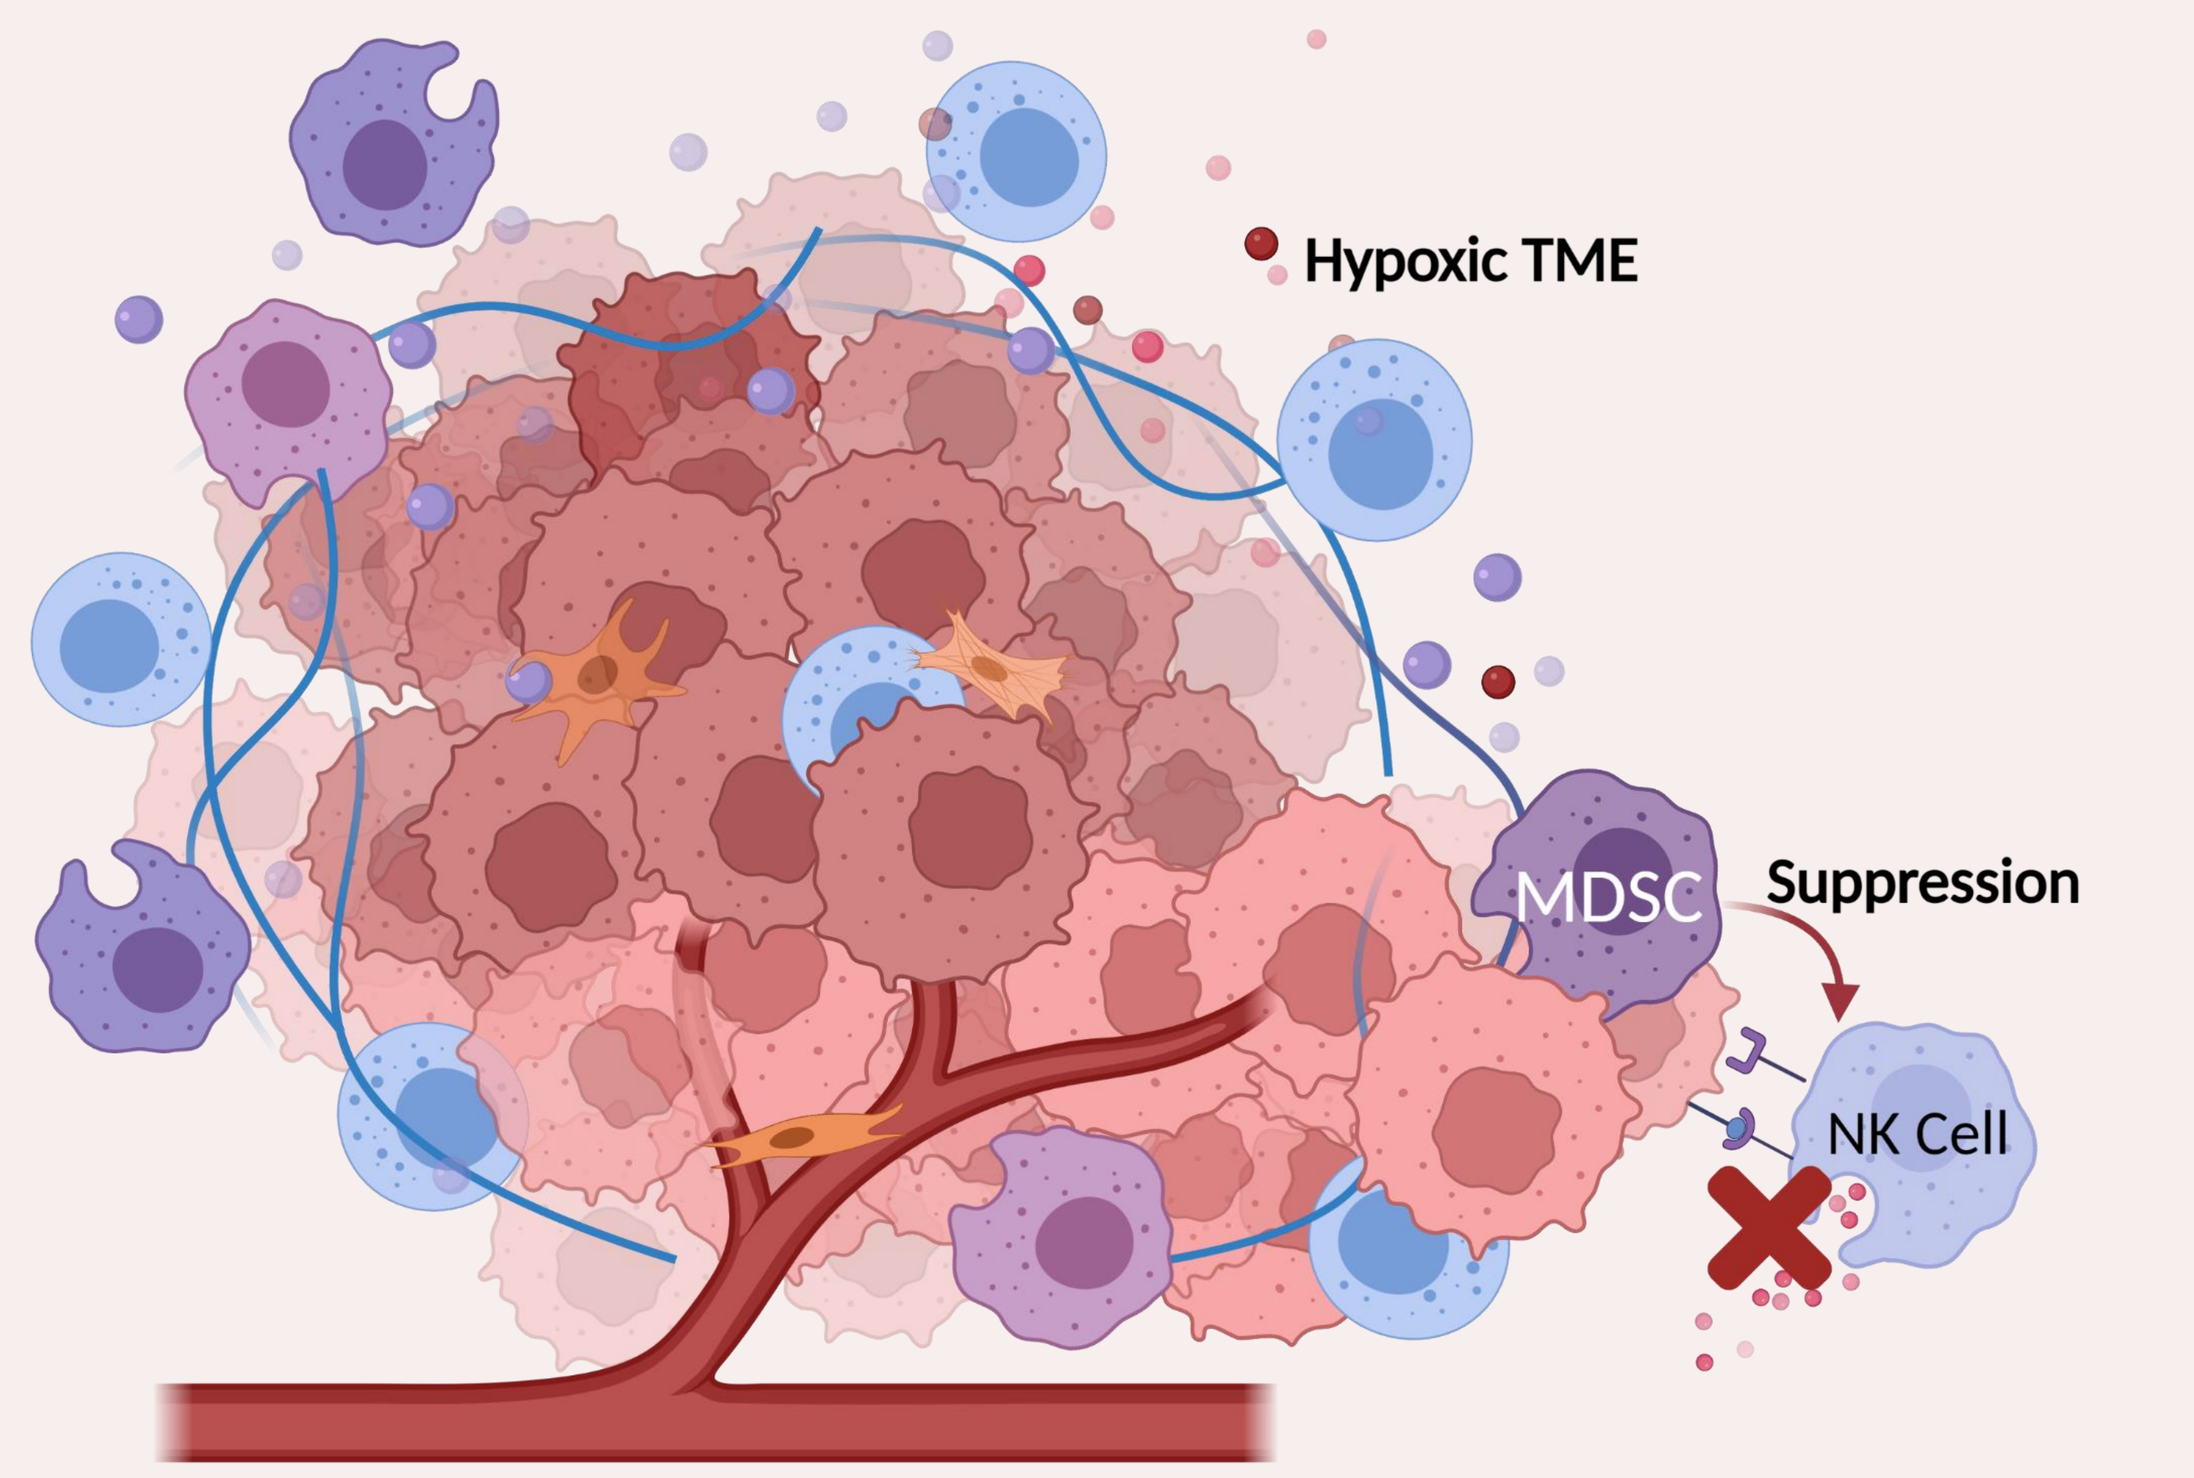
Hypoxic TME
MDSC
Suppression
NK Cell

Tri-specific Killer Engager (TriKE®)

To improve NK cell anti-tumor responses against mCRPC in the TME, we designed a novel tri-specific killer engager (TriKE) molecule that consists of:

- an arm that engages with CD16, an activating receptor of NK cells.
- an arm that binds to prostate-specific membrane antigen (PSMA) that is highly and specifically expressed on mCRPC.
- an interleukin (IL)-15 moiety that is essential for NK cell survival, proliferation, priming and motility.

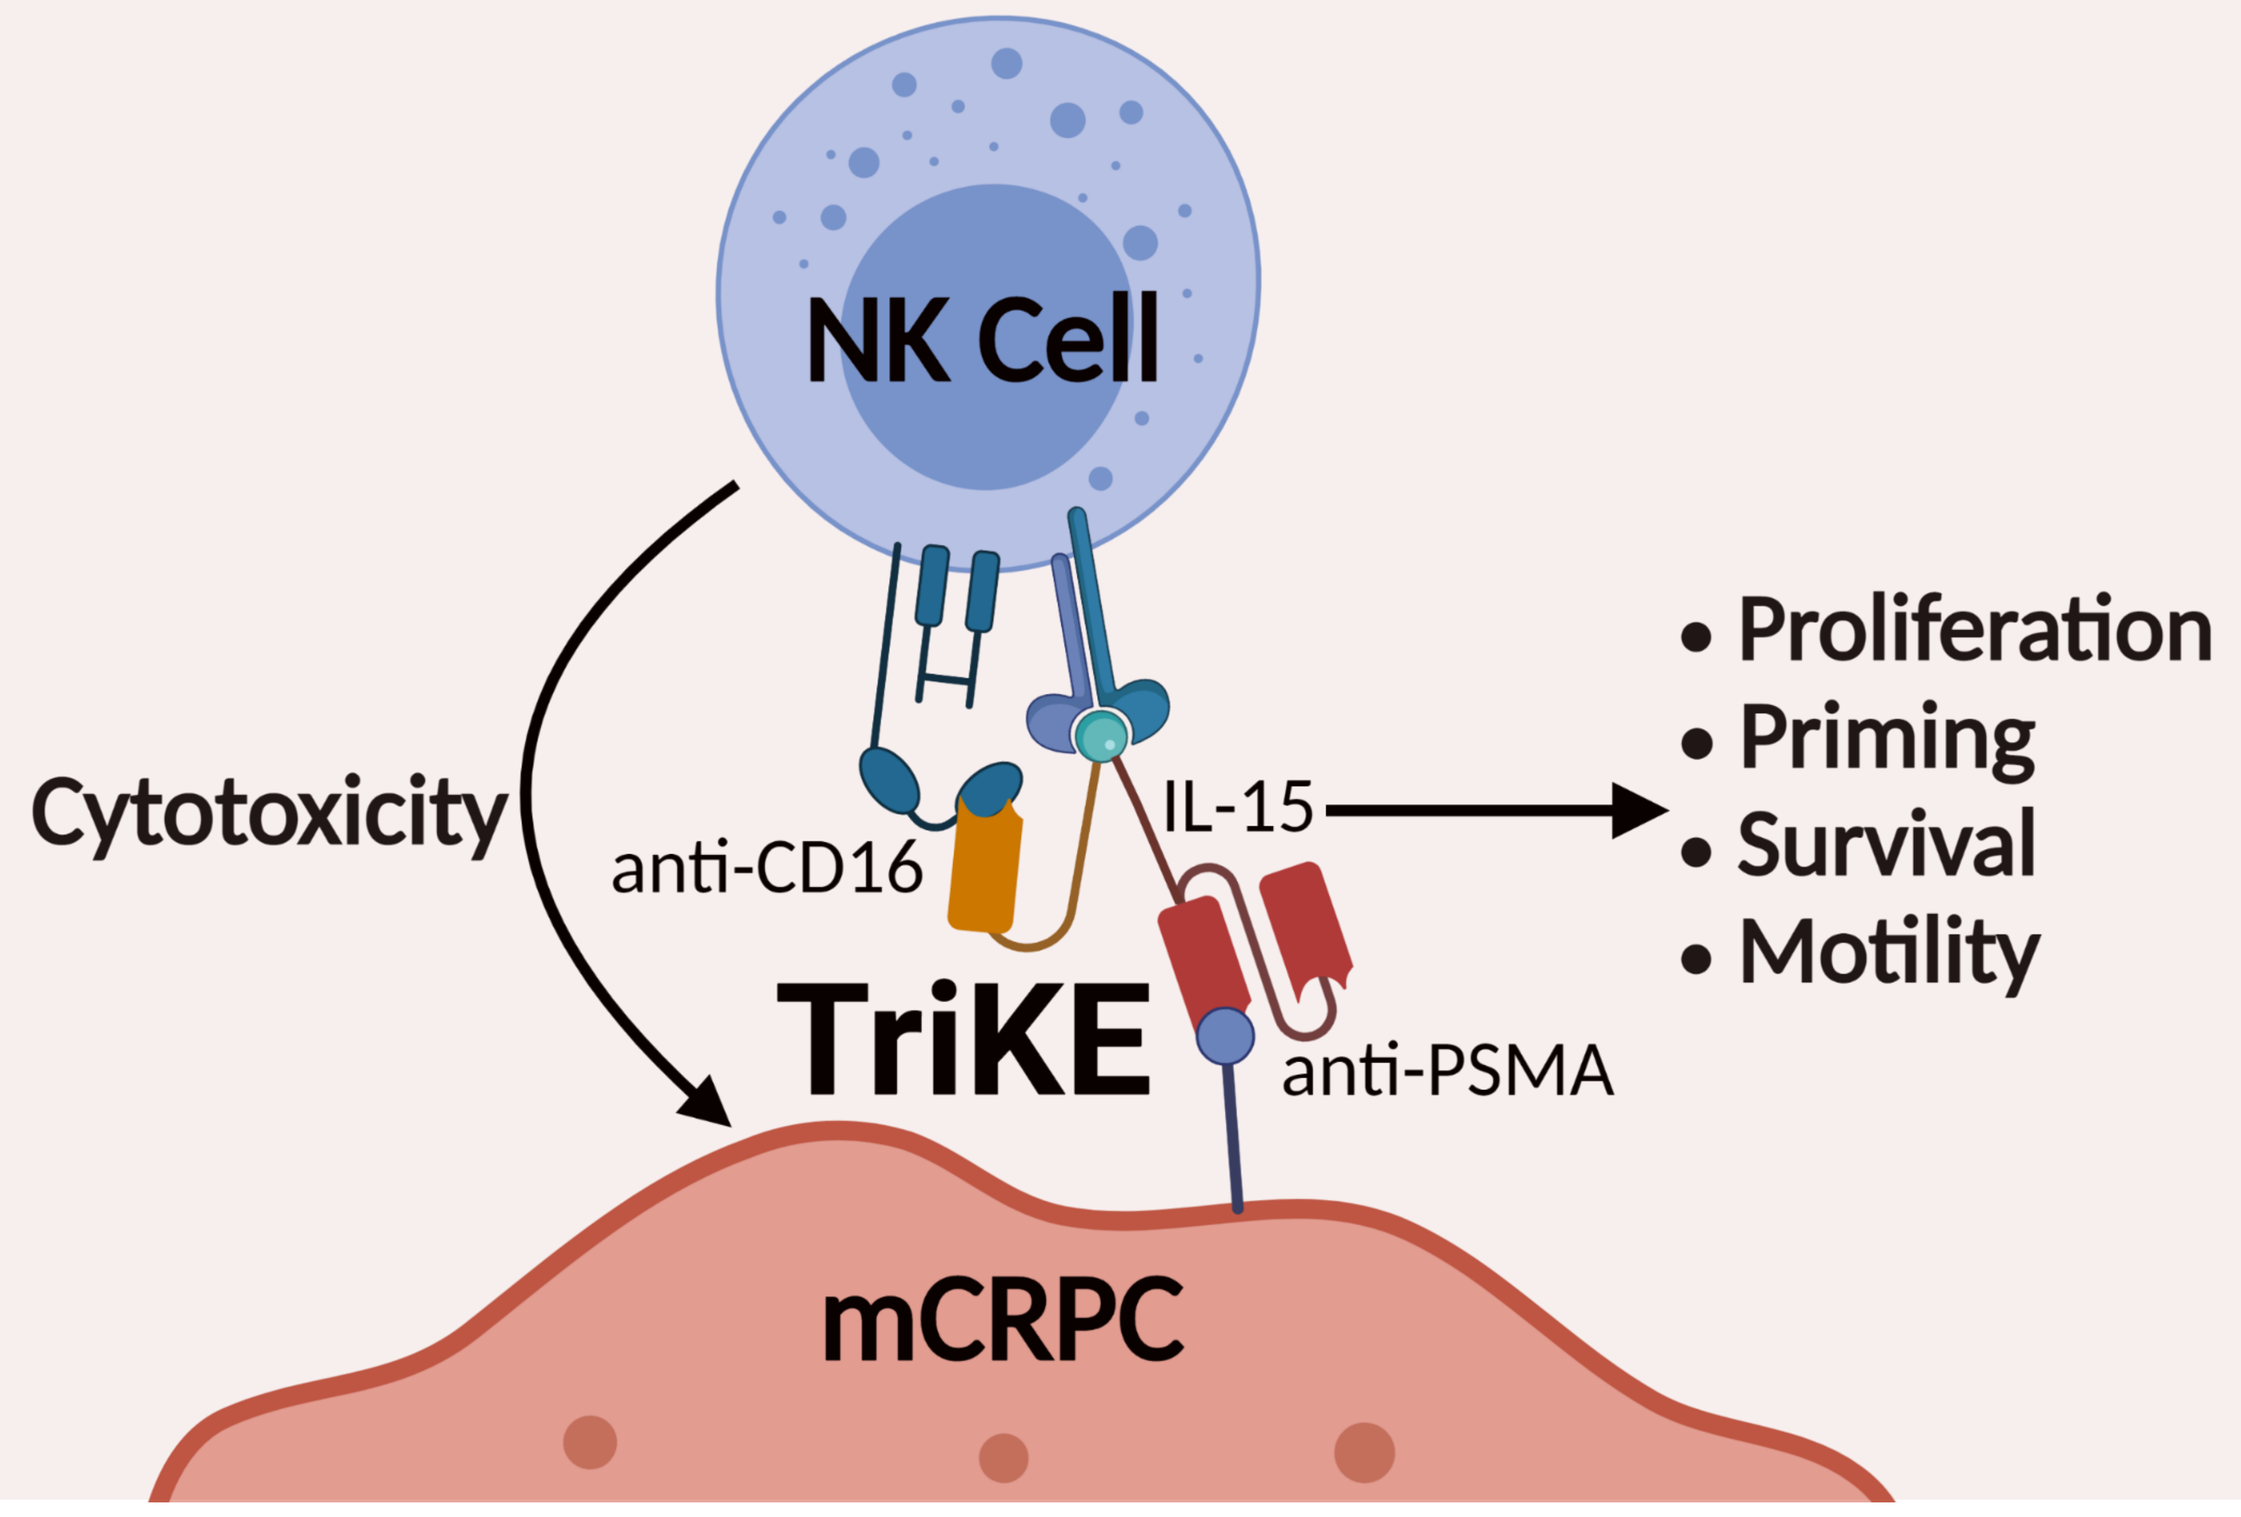
NK Cell
Cytotoxicity
anti-CD16
IL-15
TriKE
anti-PSMA
• Proliferation
• Priming
• Survival
• Motility
mCRPC

RESULTS

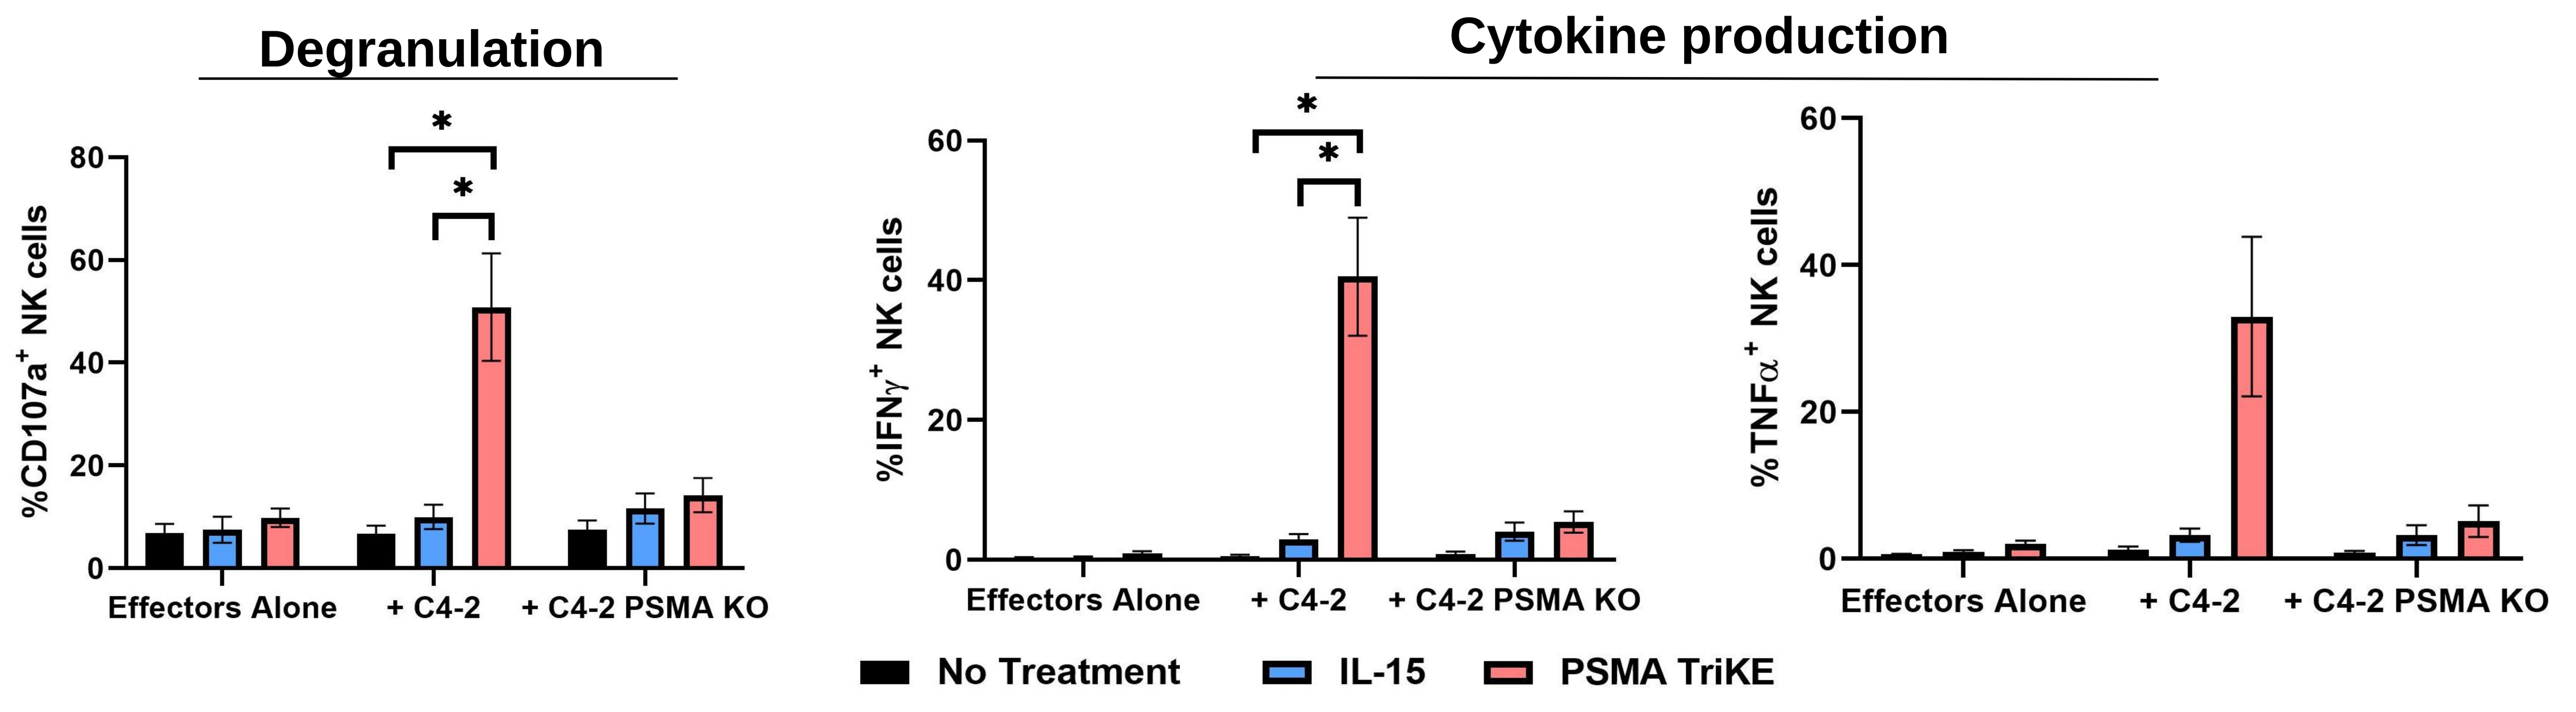
Degranulation
Cytokine production
%CD107a+ NK cells
%IFNγ+ NK cells
%TNFα+ NK cells
Effectors Alone
+ C4-2
+ C4-2 PSMA KO
No Treatment
IL-15
PSMA TriKE

Figure 1. PSMA TriKE induces specific NK cell activation against PSMA-expressing prostate cancer cell line. NK Cell degranulation (detected by presence of CD107a) and cytokine production of IFNγ and TNFα measured via flow cytometry after co-culture of peripheral blood mononuclear cells (PBMC) with PSMA+ prostate cancer cell line, C4-2, or PSMA knockout (KO) C4-2, at effector to target (E:T) ratio of 2:1 with no treatment, 3nM IL-15 or 3nM PSMA TriKE. N=4; mean ± SEM (Two-way ANOVA with Tukey's multiple comparison test); * $p < 0.05$.

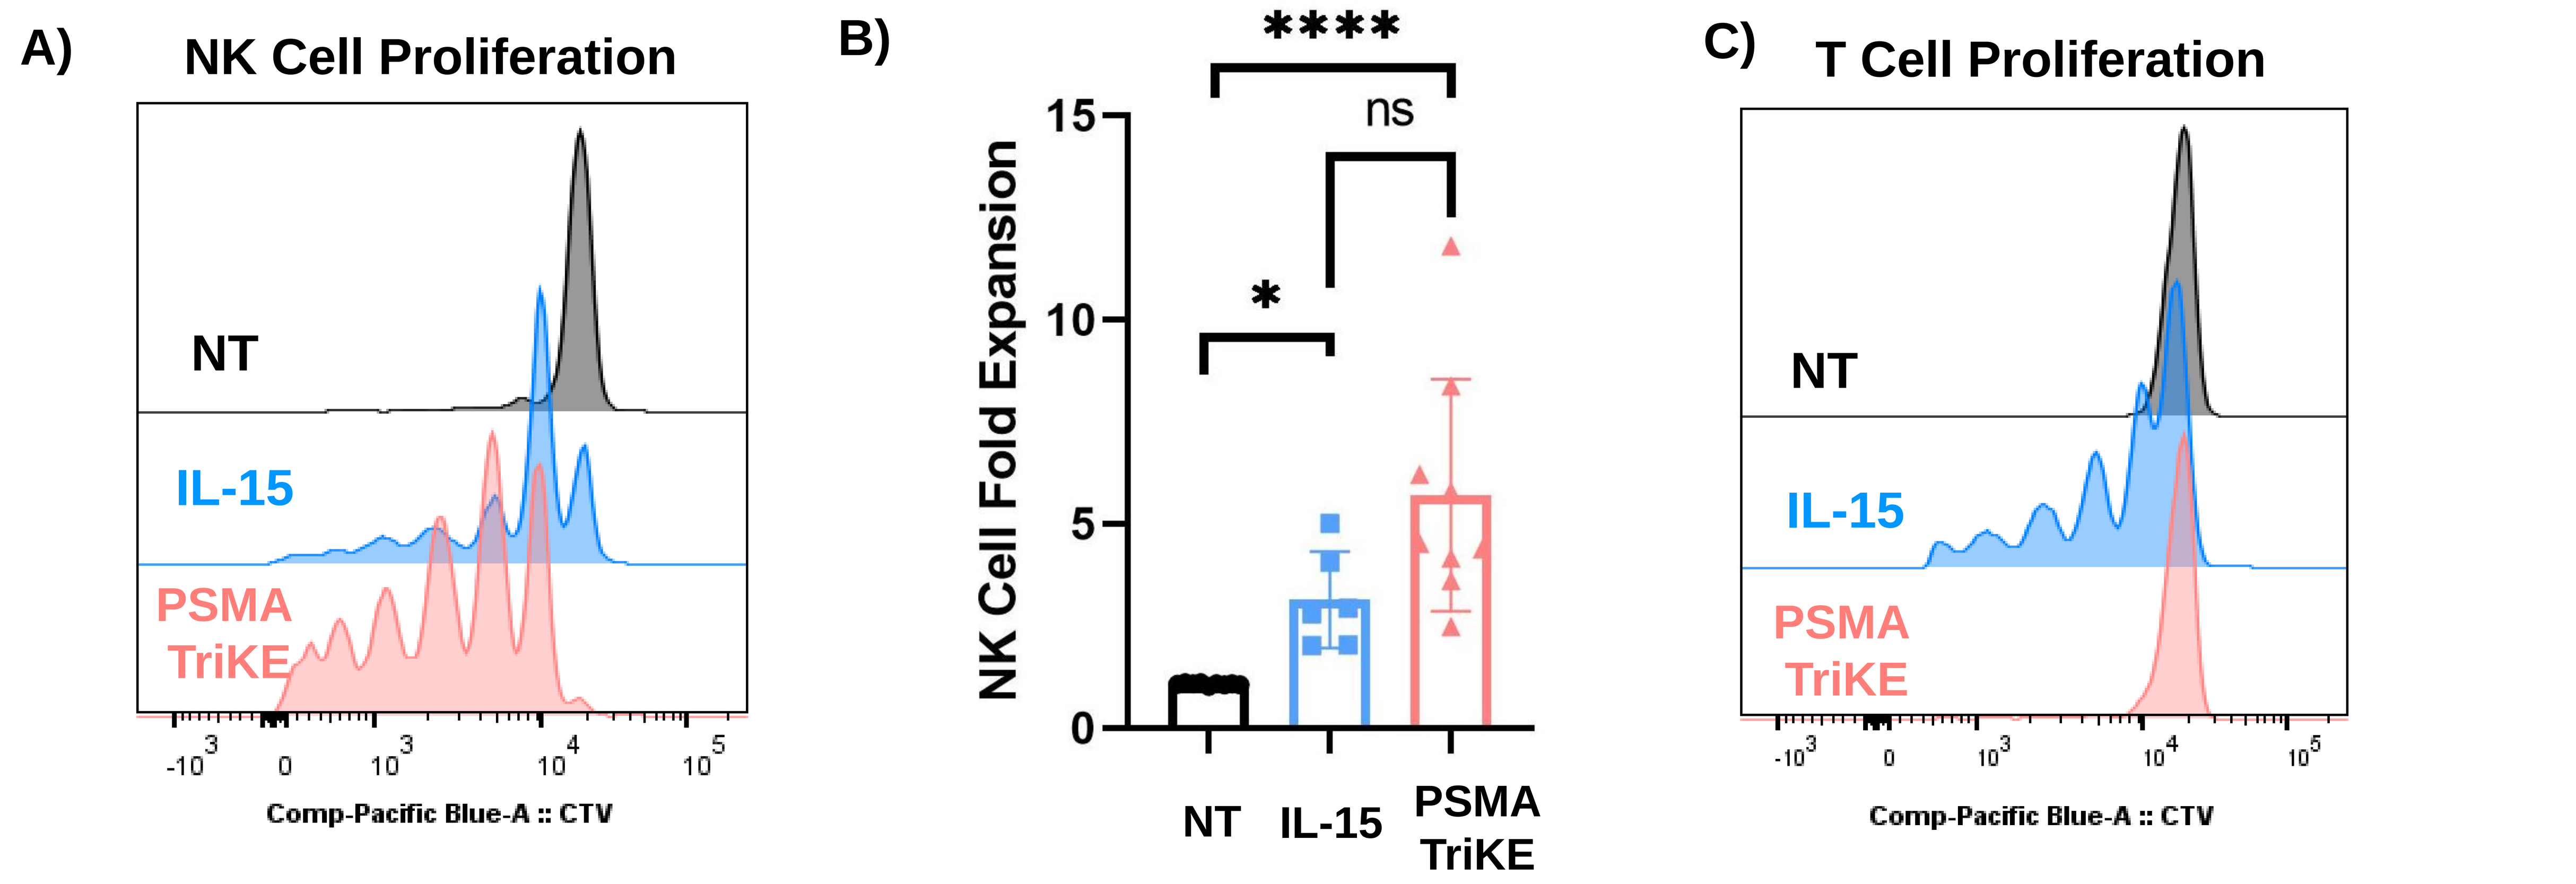
A)
NK Cell Proliferation
NT
IL-15
PSMA TriKE
Comp-Pacific Blue-A :: CTV
B)

ns
*
NK Cell Fold Expansion
NT
IL-15
PSMA TriKE
C)
T Cell Proliferation
NT
IL-15
PSMA TriKE
Comp-Pacific Blue-A :: CTV

Figure 2. PSMA TriKE preferentially enhances NK cell proliferation. A) Representative histograms of cell trace violet (CTV) dye dilution indicating proliferation of NK cells within PBMC after 7 days culture with no treatment (NT), 3nM IL-15 or 3nM PSMA TriKE. B) Cumulative results of NK cell fold expansion in 2A) with indicated treatments; N=9; mean ± SEM (Friedman with Dunn's multiple comparison test); * $p < 0.05$, **** $p< 0.0001$ C) Representative histograms of T cell proliferation within PBMC after 7 days culture with indicated treatments represented by CTV dye dilution.

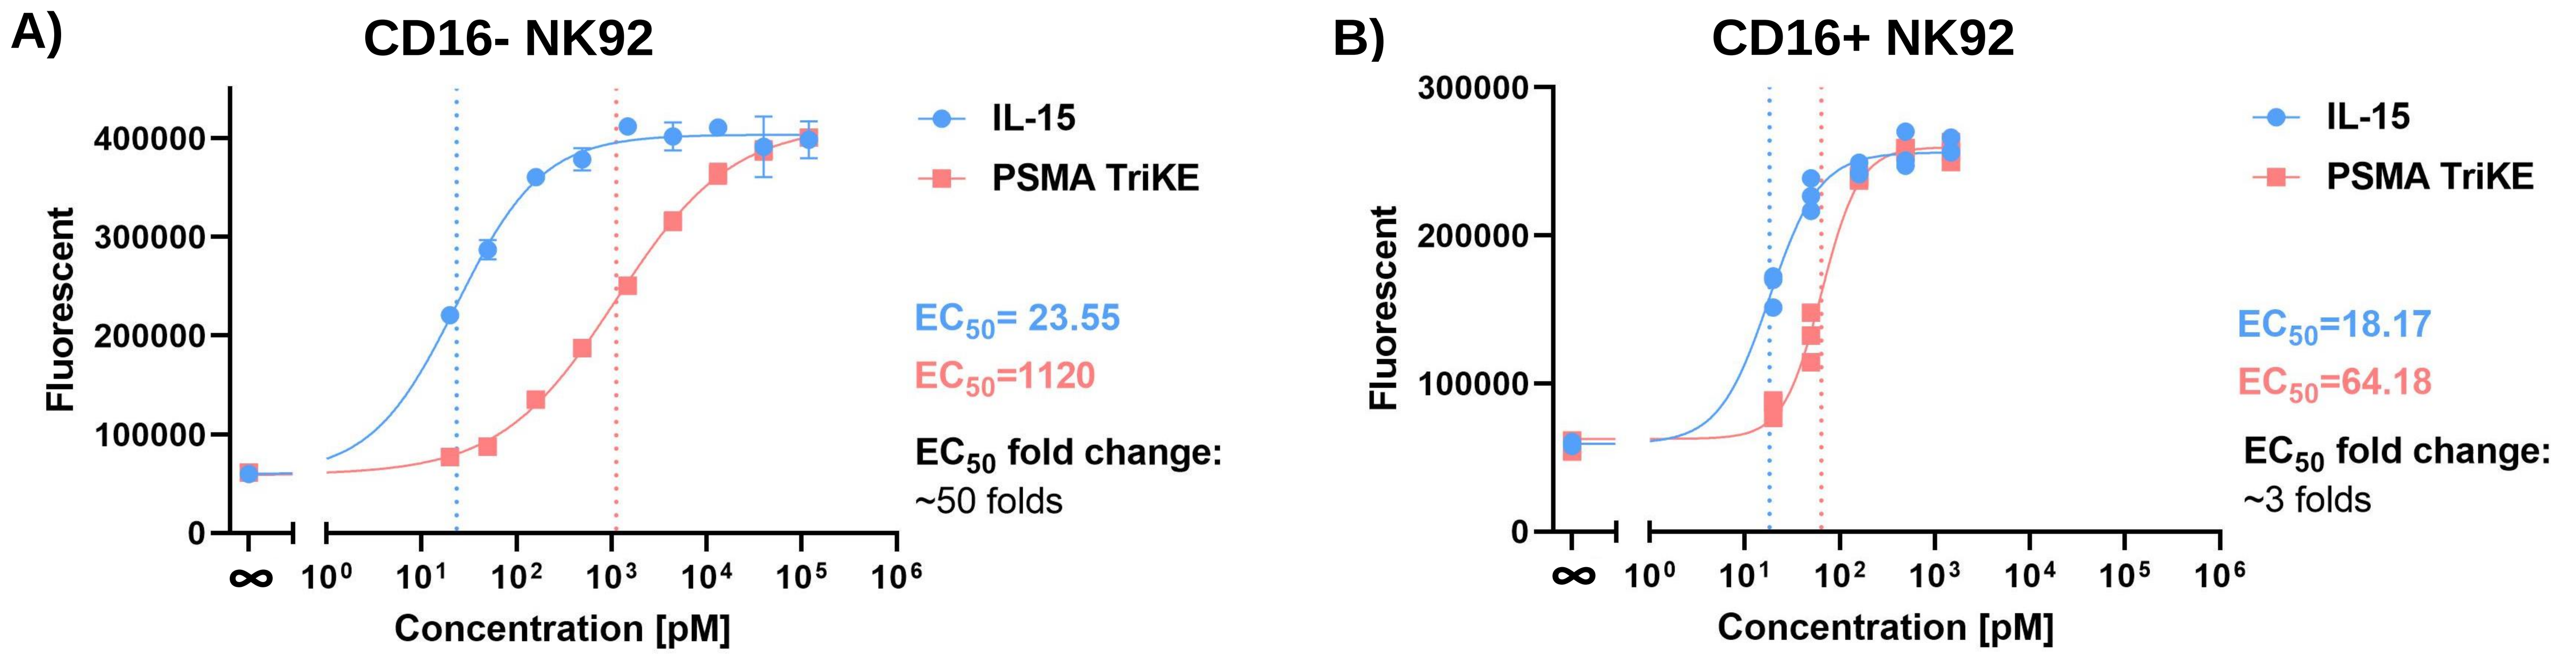
A)
CD16- NK92
IL-15
PSMA TriKE
EC50= 23.55
EC50=1120
EC50 fold change:
~50 folds
Fluorescent
Concentration [pM]
B)
CD16+ NK92
IL-15
PSMA TriKE
EC50=18.17
EC50=64.18
EC50 fold change:
~3 folds
Fluorescent
Concentration [pM]

Figure 3. CD16 is involved in the enhanced delivery of IL-15 by PSMA TriKE. Best fit curve of fluorescent emitted by Resazurin reagent that is reduced by metabolically active and viable A) CD16- NK92 or B) CD16+ NK92 after 48 hours incubation with a serially diluted range of IL-15 or PSMA TriKE; N=3.

RESULTS

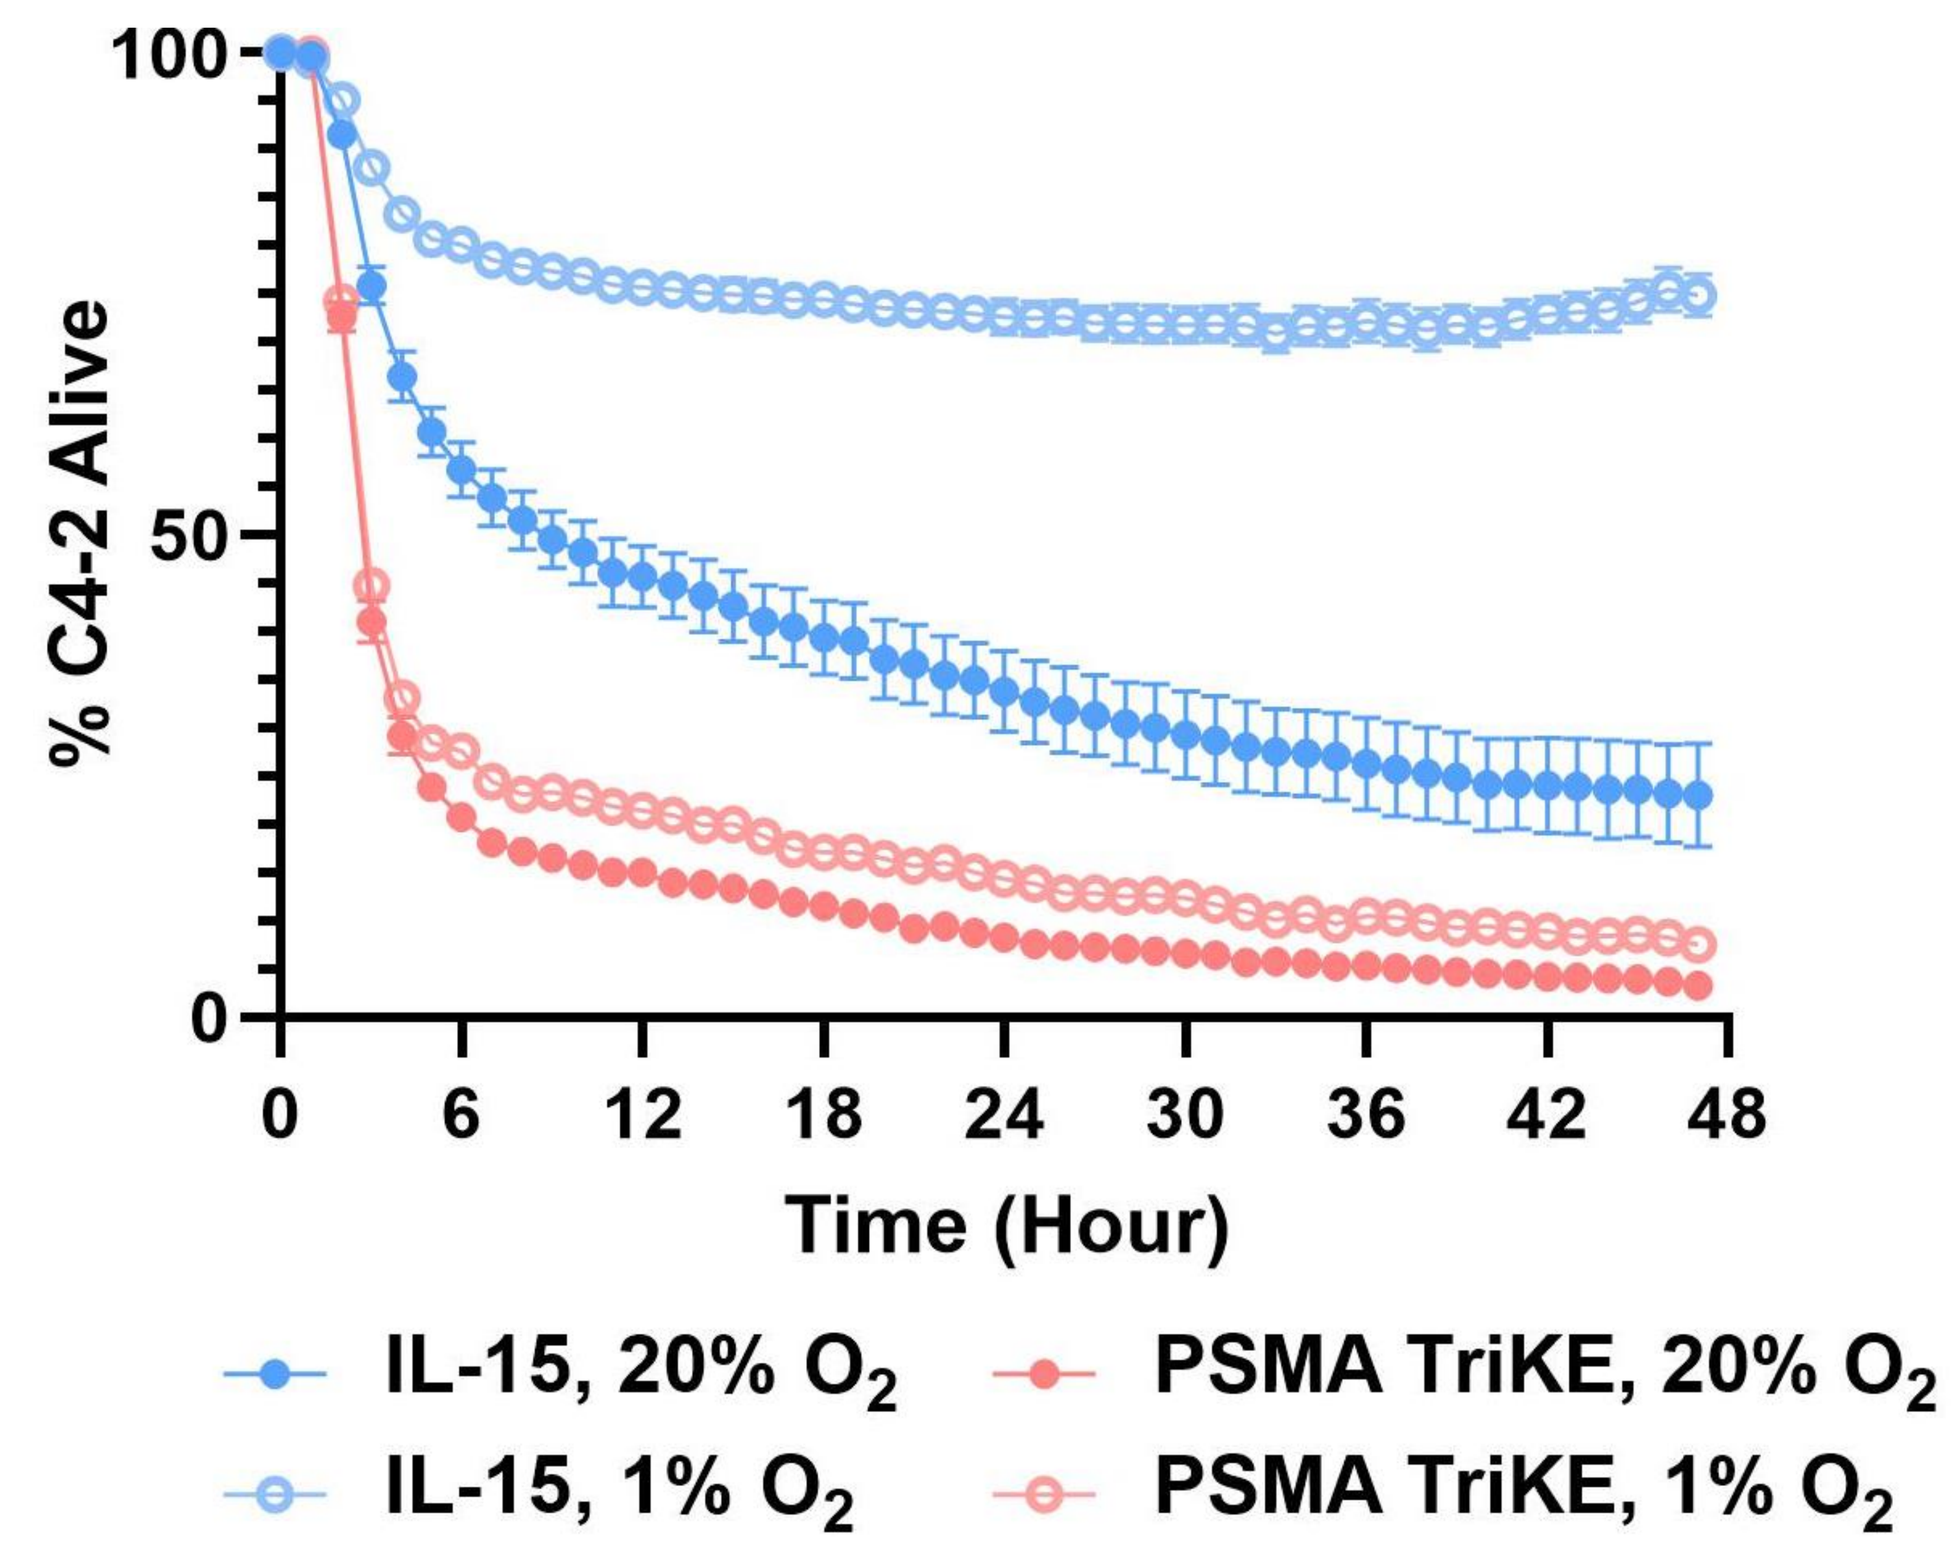
% C4-2 Alive
Time (Hour)
IL-15, 20% O2
PSMA TriKE, 20% O2
IL-15, 1% O2
PSMA TriKE, 1% O2

Figure 4. PSMA TriKE sustains NK cell cytolytic capacity after long term incubation in hypoxia. Representative IncuCyte live cell imaging assay showing the normalized percent of C4-2 alive over time. NK cells were incubated with 0.06nM IL-15 or 3nM PSMA TriKE (at equifunctional concentrations) in 20% or 1% oxygen condition for 7 days then NK cell cytotoxicity was assessed at E:T ratio of 2:1 for 48 hours; N=5.

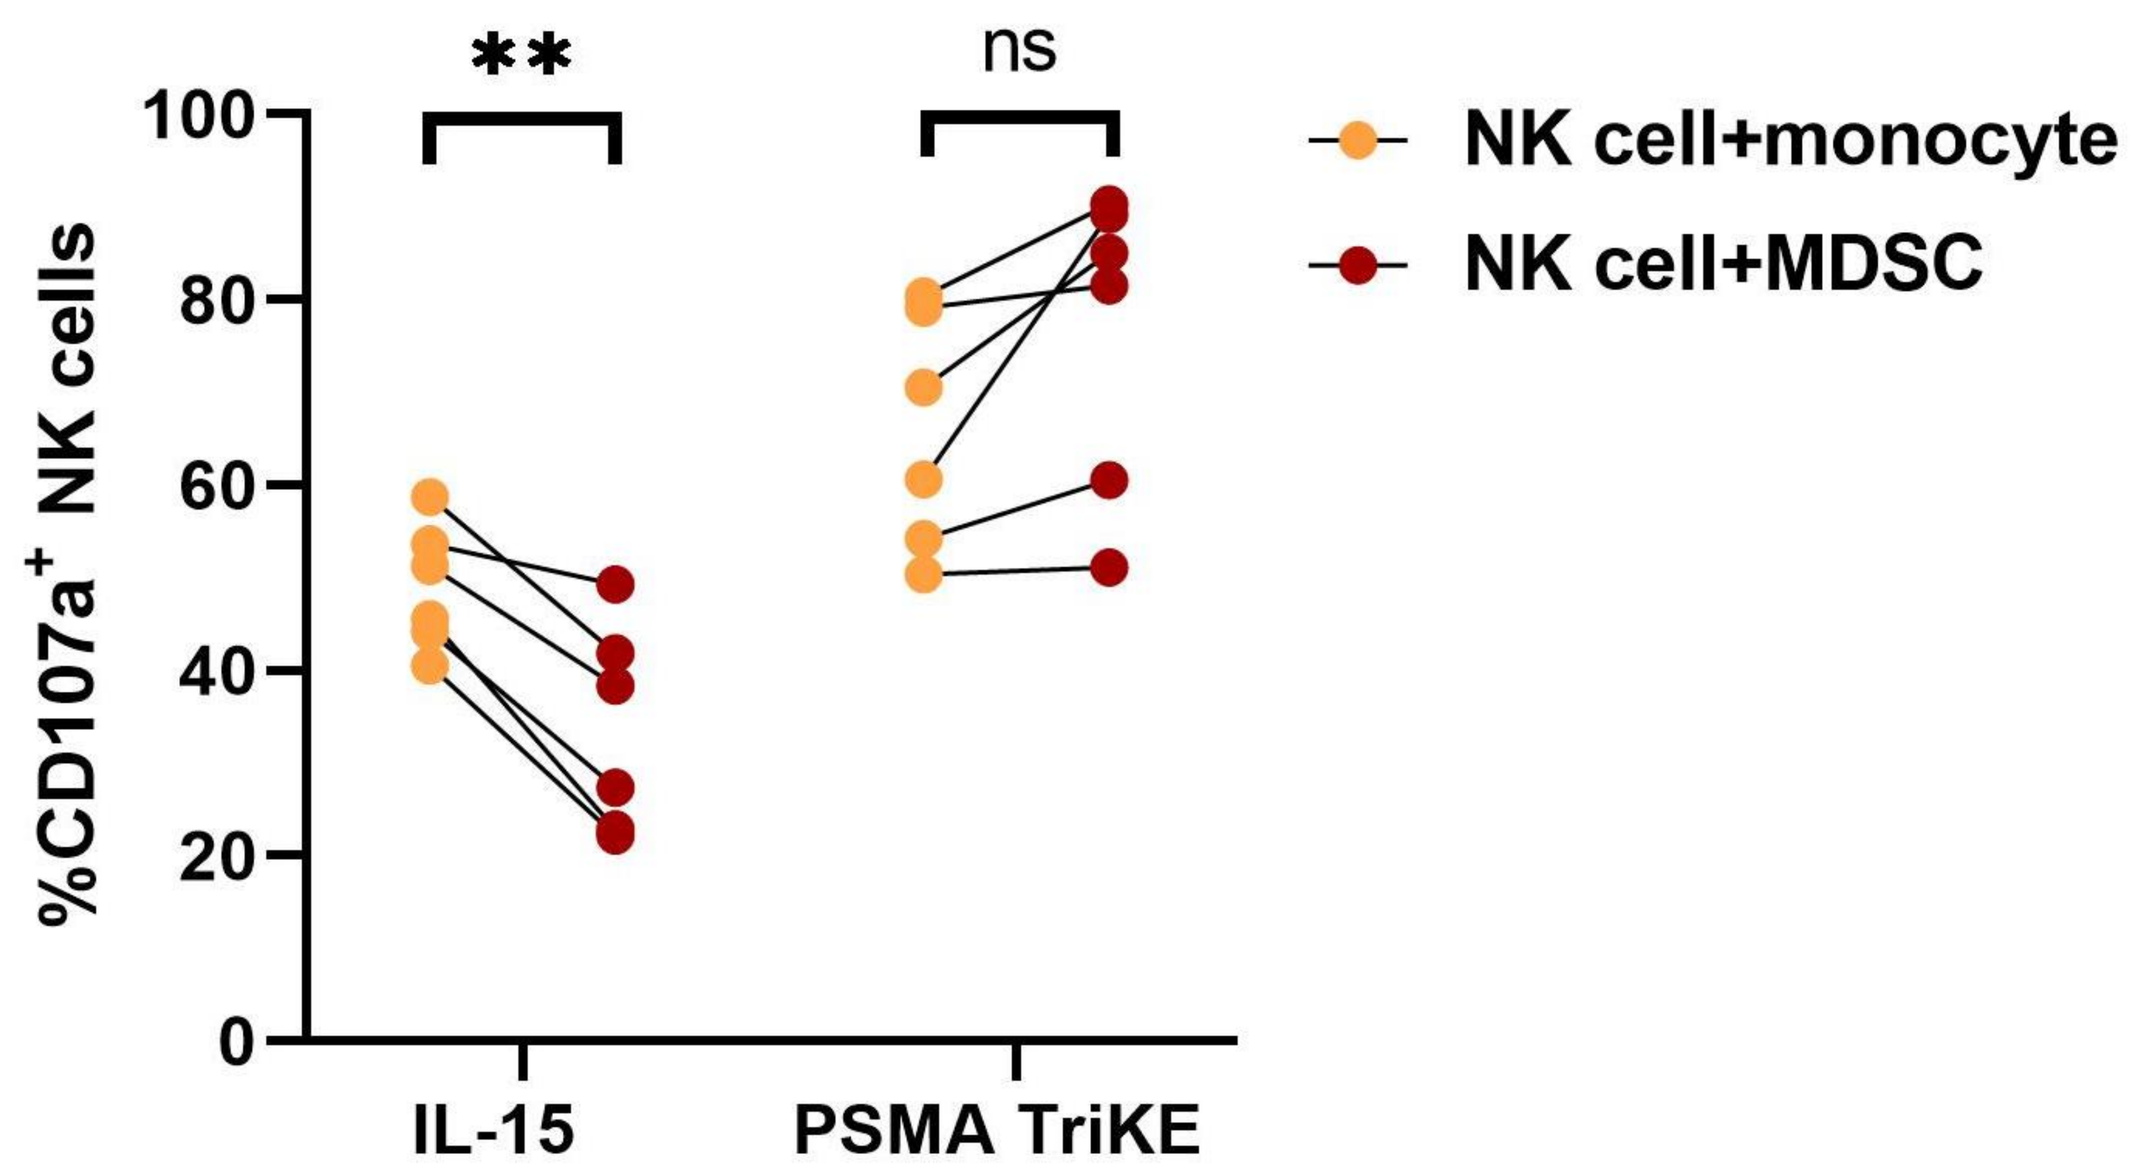
**
ns
%CD107a+ NK cells
IL-15
PSMA TriKE
NK cell+monocyte
NK cell+MDSC

Figure 5. PSMA TriKE maintains NK cell degranulation after co-culture with MDSC. NK cells were co-cultured with monocytes or cytokine-induced monocyte-derived MDSCs for 5 days with 0.06nM IL-15 or 3nM PSMA TriKE. C4-2 were used as tumor targets to induce NK cell degranulation, detected by CD107a staining. N=6 (Sidak's multiple comparisons test); ** $p < 0.01$.

CONCLUSIONS

PSMA TriKE induces specific NK cell proliferation and activation against PSMA+ tumor cells. Efficient delivery of IL-15 to NK cells by PSMA TriKE robustly relieves NK cells from suppression induced by hypoxia and MDSCs.

Acknowledgements: This work is supported by the Department of Defense (W81XWH-20-1-0659). The University Flow Cytometry Resources (UFCR), University of Minnesota, was used for all flow cytometry-based assays. Hypoxic studies were done using the AVATAR™, Xcellbio. Illustrations were made using Biorender.com.