Favourable Clinical Outcome of Two Patients with *Candida* spp. Spondylodiscitis treated with Oral Ibrexafungerp (formerly SCY-078) from the FURI Study

UNIKLINIK KÖLN

SCYNEXIS

L0033

29th ECCMID EUROPEAN CONGRESS OF CLINICAL MICROBIOLOGY AND INFECTIOUS DISEASES

Amsterdam, Netherlands
13 – 16 April 2019

Philipp Koehler[1], Oliver A. Cornely[1,2], David Angulo[3]

[1]Dept. for Internal Medicine, Excellence Center for Medical Mycology (ECMM), University of Cologne, Germany and CECAD Cluster of Excellence, University of Cologne, DE, [2]German Centre for Infection Research, Partner Site Bonn-Cologne, Cologne, Germany and Clinical Trials Centre Cologne (ZKS Köln), University of Cologne, Cologne, DE, [3]SCYNEXIS, Inc., Jersey City, NJ, USA

BACKGROUND

Fungal spondylodiscitis is a rare disease with increasing incidence due to use of immunosuppressive therapy and indwelling devices that per ESCMID-EFISG recommendation requires antifungal therapy for 6-12 months. Ibrexafungerp (SCY-078) is an orally bioavailable antifungal targeting the β-1,3-D-glucan synthase and is active against *Candida*, *Aspergillus* and *Pneumocystis*. Studies with radiolabelled ibrexafungerp in rats indicate extensive tissue distribution and bone exposures higher than blood (AUC bone:blood ratio = 1.36). A Phase 3 open-label, single-arm study of ibrexafungerp (FURI; NCT03059992) is ongoing for the treatment of patients intolerant of or with fungal disease refractory to standard antifungal therapy. We report here two cases of fungal bone and joint infection successfully treated with oral ibrexafungerp

METHODS

We report the clinical outcome of two patients with *Candida* spondylodiscitis treated with oral ibrexafungerp from the FURI study. Patients were eligible for enrollment if they had proven or probable, invasive or severe mucocutaneous candidiasis and documented evidence of failure of, intolerance to, or toxicity related to a currently approved standard-of-care antifungal treatment or could not receive approved oral antifungal options (e.g., susceptibility of the organism) and a continued IV antifungal therapy was undesirable or unfeasible due to clinical or logistical circumstances. Both subjects received a loading dose of oral ibrexafungerp 750 mg BID during the first 2 days, followed by oral ibrexafungerp 750 mg QD, for 90 days (FURI study) and subsequently received addition days of ibrexfungerp therapy under an expanded access program.

An independent Data Review Committee (DRC) provided an assessment of treatment response for 20 patients who completed oral ibrexafungerp therapy in the FURI study, including one of the Candida spondylodiscitis patients reported here. The global outcome for the 20 patients analyzed in the FURI study by the DRC is given in Table 1.

RESULTS

Table 1: Patient outcomes as determined by DRC for All Patients in the FURI Study

	Complete/Partial Response	Stable Disease	Progression of Disease	Indeterminate
All Patients (20)	11 (55%)	6 (30%)	2 (10%)	1 (5%)

RESULTS

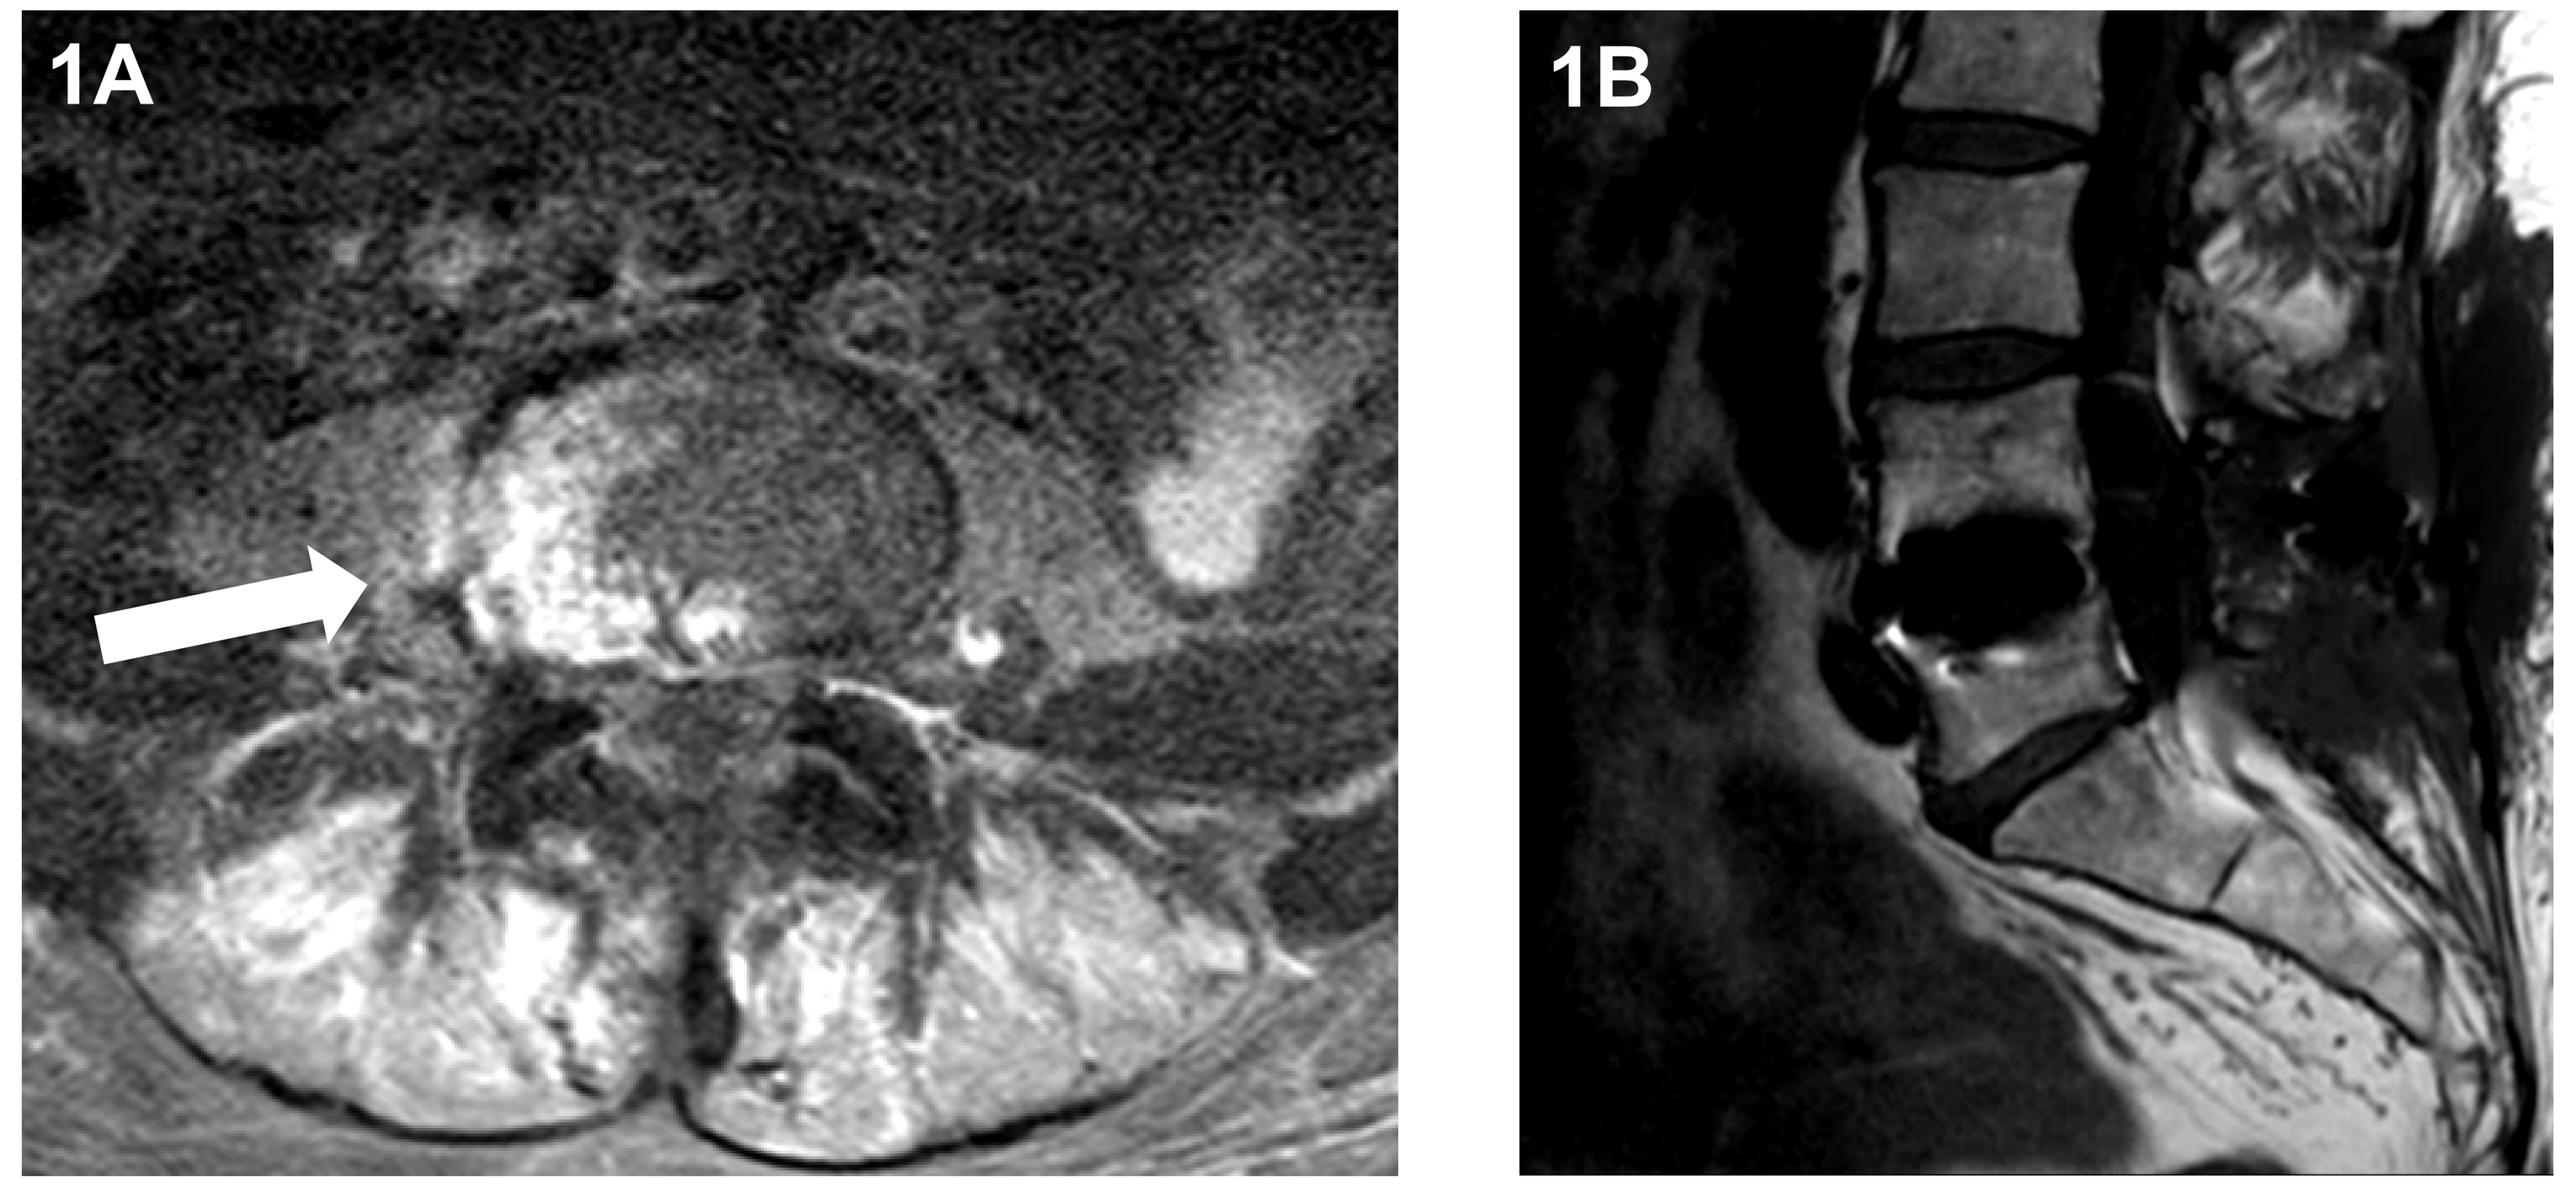
1A
1B

Patient #1: A 50 year old male with history of AML, allogeneic SCT, GVHD, and intolerant to azoles, developed *C. albicans* spondylodiscitis L4/L5 (Figure 1) five months after *C. albicans* candidaemia. The patient received posterior lumbar interbody fusion L4/L5 (PLIF) and has been treated with ibrexafungerp for 295 days (ongoing). The spondylodiscitis is considered resolved, with resolution of clinical symptoms and MRI signs of infection, and treatment is ongoing as the guideline recommended treatment duration has not yet been reached.

Figure 1: Magnetic Resonance Imaging of Patient #1.
(1A) Patient #1 axial magnetic resonance imaging (MRI-T1 SPIR) of the lumbar spine revealed abnormal signal/enhancement of the L4/L5 vertebral marrow indicative of spondylodiscitis with adjacent inflammation. **(1B)** Sagittal MRI (T1) after PLIF with cage placement. MRI follow up shows resolved inflammation.

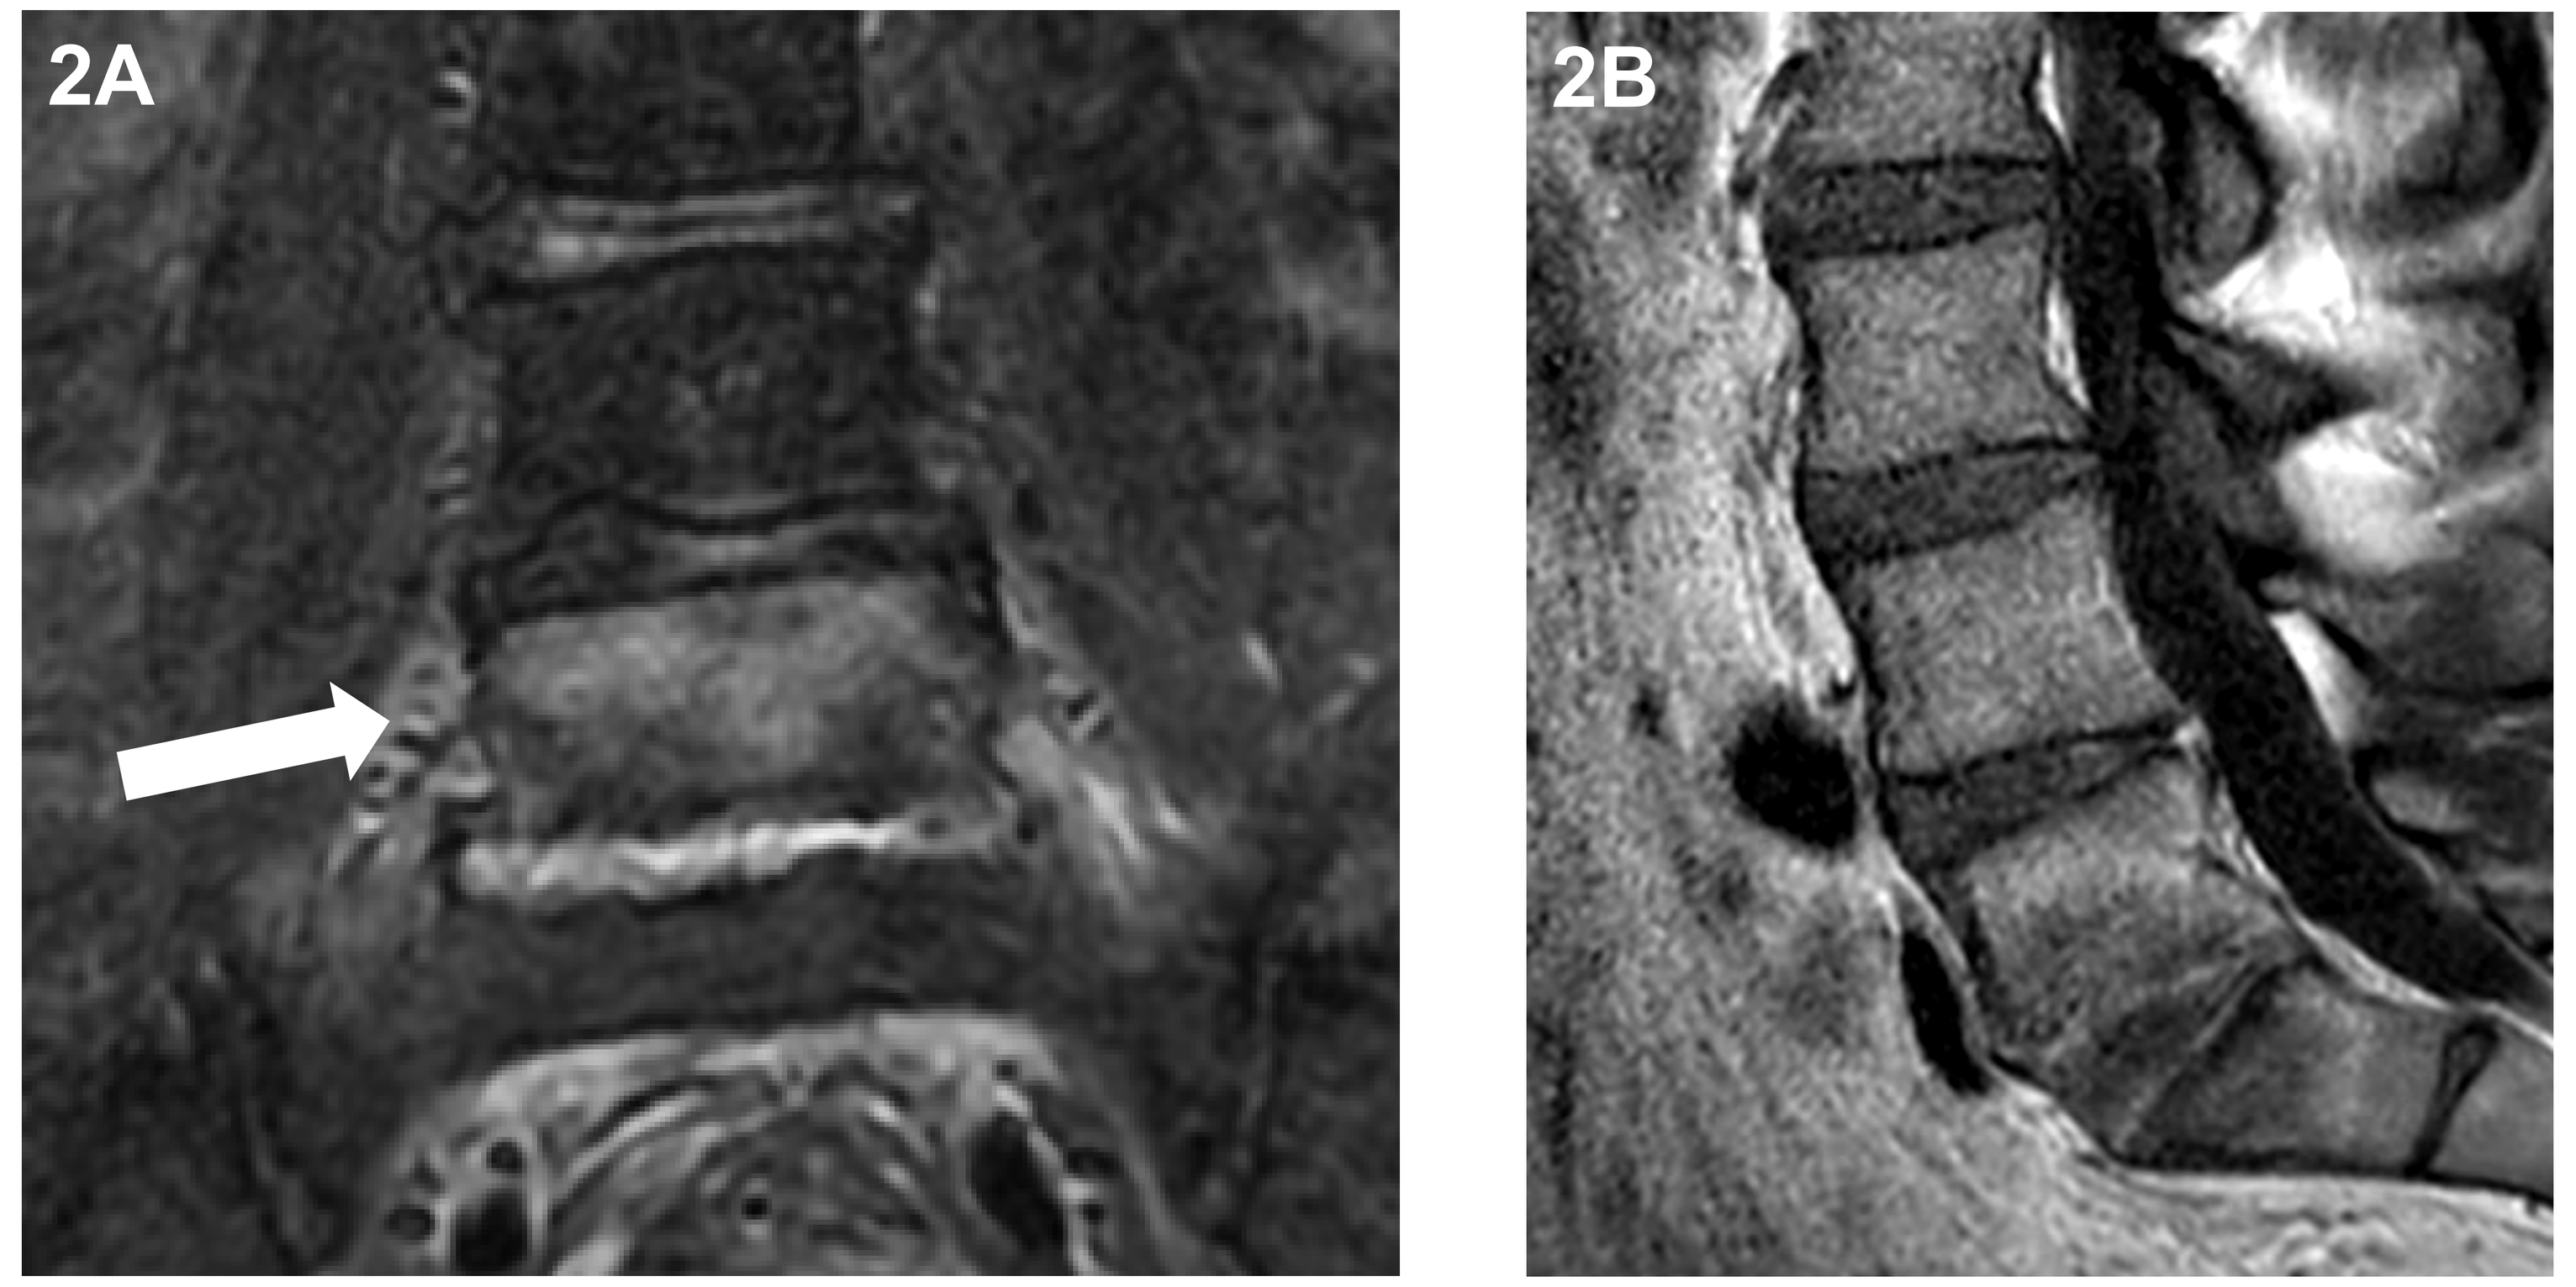
2A
2B

Patient #2: A 58 year old male with relapsed bladder cancer after cystectomy and ileum conduit developed *C. tropicalis* spondylodiscitis L5/S1 (Figure 2) three months after *C. tropicalis* fungemia. The patient was enrolled into the FURI study due to refractoriness to standard therapy, received PLIF L5/S1 and ibrexafungerp for 106 days (ongoing). The spondylodiscitis is considered resolved, with resolution of clinical symptoms and MRI signs of infection, and treatment is ongoing as the guideline recommended treatment duration has not yet been reached.

Figure 2: Magnetic Resonance Imaging of Patient #2.
(2A) Coronal MRI (T2 Flex) of the lumbar spine revealed abnormal signal/enhancement of the L5/S1 vertebral marrow including intervertebral space indicative of spondylodiscitis with adjacent inflammation. **(2B)** Sagittal MRI (T1) after PLIF. MRI follow up shows resolved inflammation.

Ibrexafungerp was well-tolerated in both patients. Possible study drug related adverse events in both patients were diarrhoea (AE Grade 2), flatulence (G1) and nausea (G1).

CONCLUSIONS

Ibrexafungerp is a novel antifungal agent active against *Candida* species. An advantage is the oral availability, which allows prolonged therapy for bone infections in the outpatient setting. Ibrexafungerp shows pre-clinical data supporting bone penetration and limited, but promising clinical data for its use in bone infections. Further investigation is warranted.

For additional information, contact info@scynexis.com