Population Pharmacokinetic Modeling and Exposure-Response Analysis of Probuphine® Implants in Opioid Use Disorder Subjects

Olivier Barriere[1], Nathalie H. Gosselin[1], Nada Farhat[1], Markus Jerling[2], Sunil Sreedharan[3]

[1]Certara Strategic Consulting, Princeton, NJ, USA; [2]Markus Jerling Consulting AB, Stavgårdsgatan, Bromma, Sweden; [3]Titan Pharmaceutical, Inc., South San Francisco, CA, USA.

CERTARA | STRATEGIC CONSULTING

- Population PK model with two depot compartments: **initial transient quick-depot followed by a sustained slow-depot**
- **Emax model with ceiling on maximum effect of opioid negative urines** in treated patients (*plasma buprenorphine* EC_{50} *= 0.181 ng/mL*)
- **Average buprenorphine concentration** over Weeks 1-24 for **4 implants** (*Cavg' = 0.638 ng/mL*) **would result in 77.9% of Emax**
- **Proportion (>30% or >50%) of opioid-negative urines** from Weeks 1-24 **increased with plasma buprenorphine Cavg' (p<0.05)**, with plateau in maximum response occurring between *0.56 – 0.85 ng/mL*

Background & Objectives

The use of buprenorphine (BPN) for the treatment of opioid dependence is well established and accepted as a safe and useful treatment for this disease. BPN is a partial agonist at mu-opioid receptors (MORs) and an antagonist at kappa-opioid receptors (KORs); therefore, a ceiling or plateau effect is observed whereby high doses of BPN are less likely to cause increased euphoria or significant complications upon overdose.

Probuphine® (buprenorphine) implant, a six-month maintenance treatment for opioid-use disorder (OUD) in eligible patients is approved by the US Food and Drug Administration (*Probuphine Prescribing Information, 2018*), Health Canada, and the European Medicines Agency.

The specific objectives for this analysis were to:

- Model the release of BPN following administration of sublingual (SL) BPN or Probuphine implants in opioid-dependent subjects to characterize the population pharmacokinetics (PK) parameters.
- Perform Monte-Carlo simulations to predict PK exposure levels after SL BPN administration or insertion of Probuphine implants.
- Conduct exposure-response (E-R) analysis for the relationship between plasma BPN and percentage of opioid negative urines with self-reported use in randomized, double-blinded, placebo-controlled Probuphine Phase 3 studies.

Methods

Clinical Trials

The population PK analysis was based on final clinical data in opioid-dependent subjects from five Phase 3 studies (two randomized, double-blinded, placebo-controlled studies [PRO-805 and PRO-806] and their respective open-label extension studies [PRO-807 and PRO-811] (*Ling, 2010; Rosenthal, 2013*), as well as the relative bioavailability open-label PK study, [PRO-810]. Exposure-response pharmacodynamics (PD) analysis was performed on data from studies PRO-805 and PRO-806.

PK Modeling Approach and Simulations

Nonlinear mixed-effect modeling was used to assess the concentration-time profiles of BPN in opioid-dependent adults. Covariate analysis was conducted based on stepwise forward addition and backward elimination procedure.

BPN profiles were simulated during 24 weeks for the two dosing regimens: i) 4 Probuphine implants (each containing 80 mg BPN hydrochloride), ii) 4 Probuphine implants with a fifth implant inserted at Week 12.

Exposure-Response Analysis

To perform the exposure-response analysis, average BPN (Cavg') were derived from individual BPN concentrations observed at Weeks 1, 4, 8, 12, 16, 20 and 24 following Probuphine implantation in studies PRO-805 and PRO-806, and were merged with percentages of opioid-negative urines with self-reported use from treatment Weeks 1-24.

Categories of responders were defined as the proportion (>50% or >30%) of opioid-negative urines from Weeks 1-24.

Results & Conclusions

A total of 363 OUD subjects treated with SL BPN doses and/or 4 Probuphine implants were included in the population PK analysis. BPN released from Probuphine implants was best characterized by a model including two depot compartments with 2.6% of absorbed implant dose being released from transient quick-release depot and 97.4% being released from the sustained slow-release depot (Figure 1). **The contribution of the sustained slow-release phase to the overall absorbed dose for the 6 month treatment duration is, therefore, greater than that of the quick-release phase.**

The rate of quick-release absorption of BPN following Probuphine implantation is 0.0668 h^{-1} with an absorption lag time of 5.8 h, and the rate of slow-release absorption is 1.19×10^{-4} h^{-1} with an absorption lag time of 9.0 h. Only the effects of body weight on apparent clearance and body mass index (BMI) on the first-order rate of slow absorption were statistically significant. Typical values of CL/F and Vc/F for BPN were 45.2 L/h and 96.0 L, respectively (from PRO-810 PK data). Based on the simulations, the concentrations were higher after the insertion of a fifth implant at Week 12 of treatment with 4 Probuphine implants (~25% greater) (Figure 2).

The relationship between plasma BPN Cavg' and percentage of opioid negative urines (Treatment Weeks 1-24) was modeled using linear and Emax models. A simple Emax model without intercept was found to provide the best fit of the data. The model indicates that **77.9% of theoretical maximum possible effect is achieved at plasma BPN of 0.638 ng/mL (Cavg' Weeks 1-24 for 4 Probuphine implants).** Noticeably, there was a **plateau in the response; higher values of plasma BPN did not significantly enhance efficacy** in this Emax model. No impact of demographic characteristics was observed between Cavg' of plasma BPN and opioid-negative urines (Weeks 1-24) (Figure 3).

A linear increase in logit of the probability of responders (proportion of opioid-negative urines from Weeks 1-24 that were >30% and >50%) with BPN Cavg' was estimated by logistic regression (p<0.05). For both endpoints, similar proportions of responders were observed on quartiles 2, 3, and 4 (31.1% vs 34.6% vs 32.7 % based on percentage >50%; and 44.7% vs 44.2% vs 45.2% based on percentage >30%), indicating a **plateau in the efficacy occurring between plasma BPN concentrations 0.56 – 0.85 ng/mL**.

Higher plasma BPN concentrations did not significantly increase the proportion of responders (Figure 4).

Discussion

The plasma BPN Cavg' for Treatment Weeks 1-24 for 4 Probuphine implants is 0.638 ng/mL, which is within the plateau seen for efficacy at concentrations of 0.56 – 0.85 ng/mL in the Probuphine Emax model. This range in maximum efficacy is similar to published data in adults which showed minimal withdrawal symptoms at plasma concentrations >0.7 ng/mL (*Kuhlman, 1998*) and EC_{50} of 0.67 ng/mL (*Laffont, 2016*).

The pharmacology of BPN and its major metabolite, norbuprenorphine (nBPN), is complex with interactions at **all of the opioid receptor subtypes** and not just at MORs. Intra-venous administration of nBPN decreased the respiratory rate in rats, whereas BPN had no effect at the same concentration (*Ohtani, 1997*). In our clinical studies the **plasma exposure ratio of nBPN:BPN with 4 Probuphine implants was 0.5:1 compared to 2:1 with 16 mg SL BPN**. Plasma BPN levels required to attain the 95th percentile of 70% occupancy for only MORs (≥2–3 ng/mL; *Haight, 2019*) are significantly above the range of maximum efficacy that we have seen in our studies, and would result in a greater exposure to the active metabolite, nBPN.

Our goal for BPN treatment of OUD is to maintain the maximum clinical benefit of BPN while keeping active metabolite levels as low as possible. Following Probuphine treatment, average BPN levels of 0.638 ng/mL (Weeks 1-24) are in the range of maximum efficacy, with relatively low levels of metabolism to nBPN observed. Further, these BPN levels delivered by Probuphine in the treatment of OUD provide significant clinical benefits, as demonstrated in multiple Phase 3 clinical studies.

References

Probuphine® (buprenorphine implant for subdermal administration CIII) prescribing information (2018). Titan Pharmaceuticals Inc.

Ling et al. (2010) JAMA, 304(14):1576-1583.

Rosenthal et al. (2013) Addiction, 108(12):2141-2149.

Kuhlman et al. (1998) Addiction, 93:549–559.

Laffont et al. (2016) J. Clin. Pharmacol., 56:806–815.

Ohtani et al. (1997) J Pharmacol. Exp. Ther., 281(1):428–33.

Haight et al. (2019) Lancet, 393(10173):778-790.

Figures

Figure 1. Schematic Representation of Two-Compartment Structural PK Model for Plasma BPN

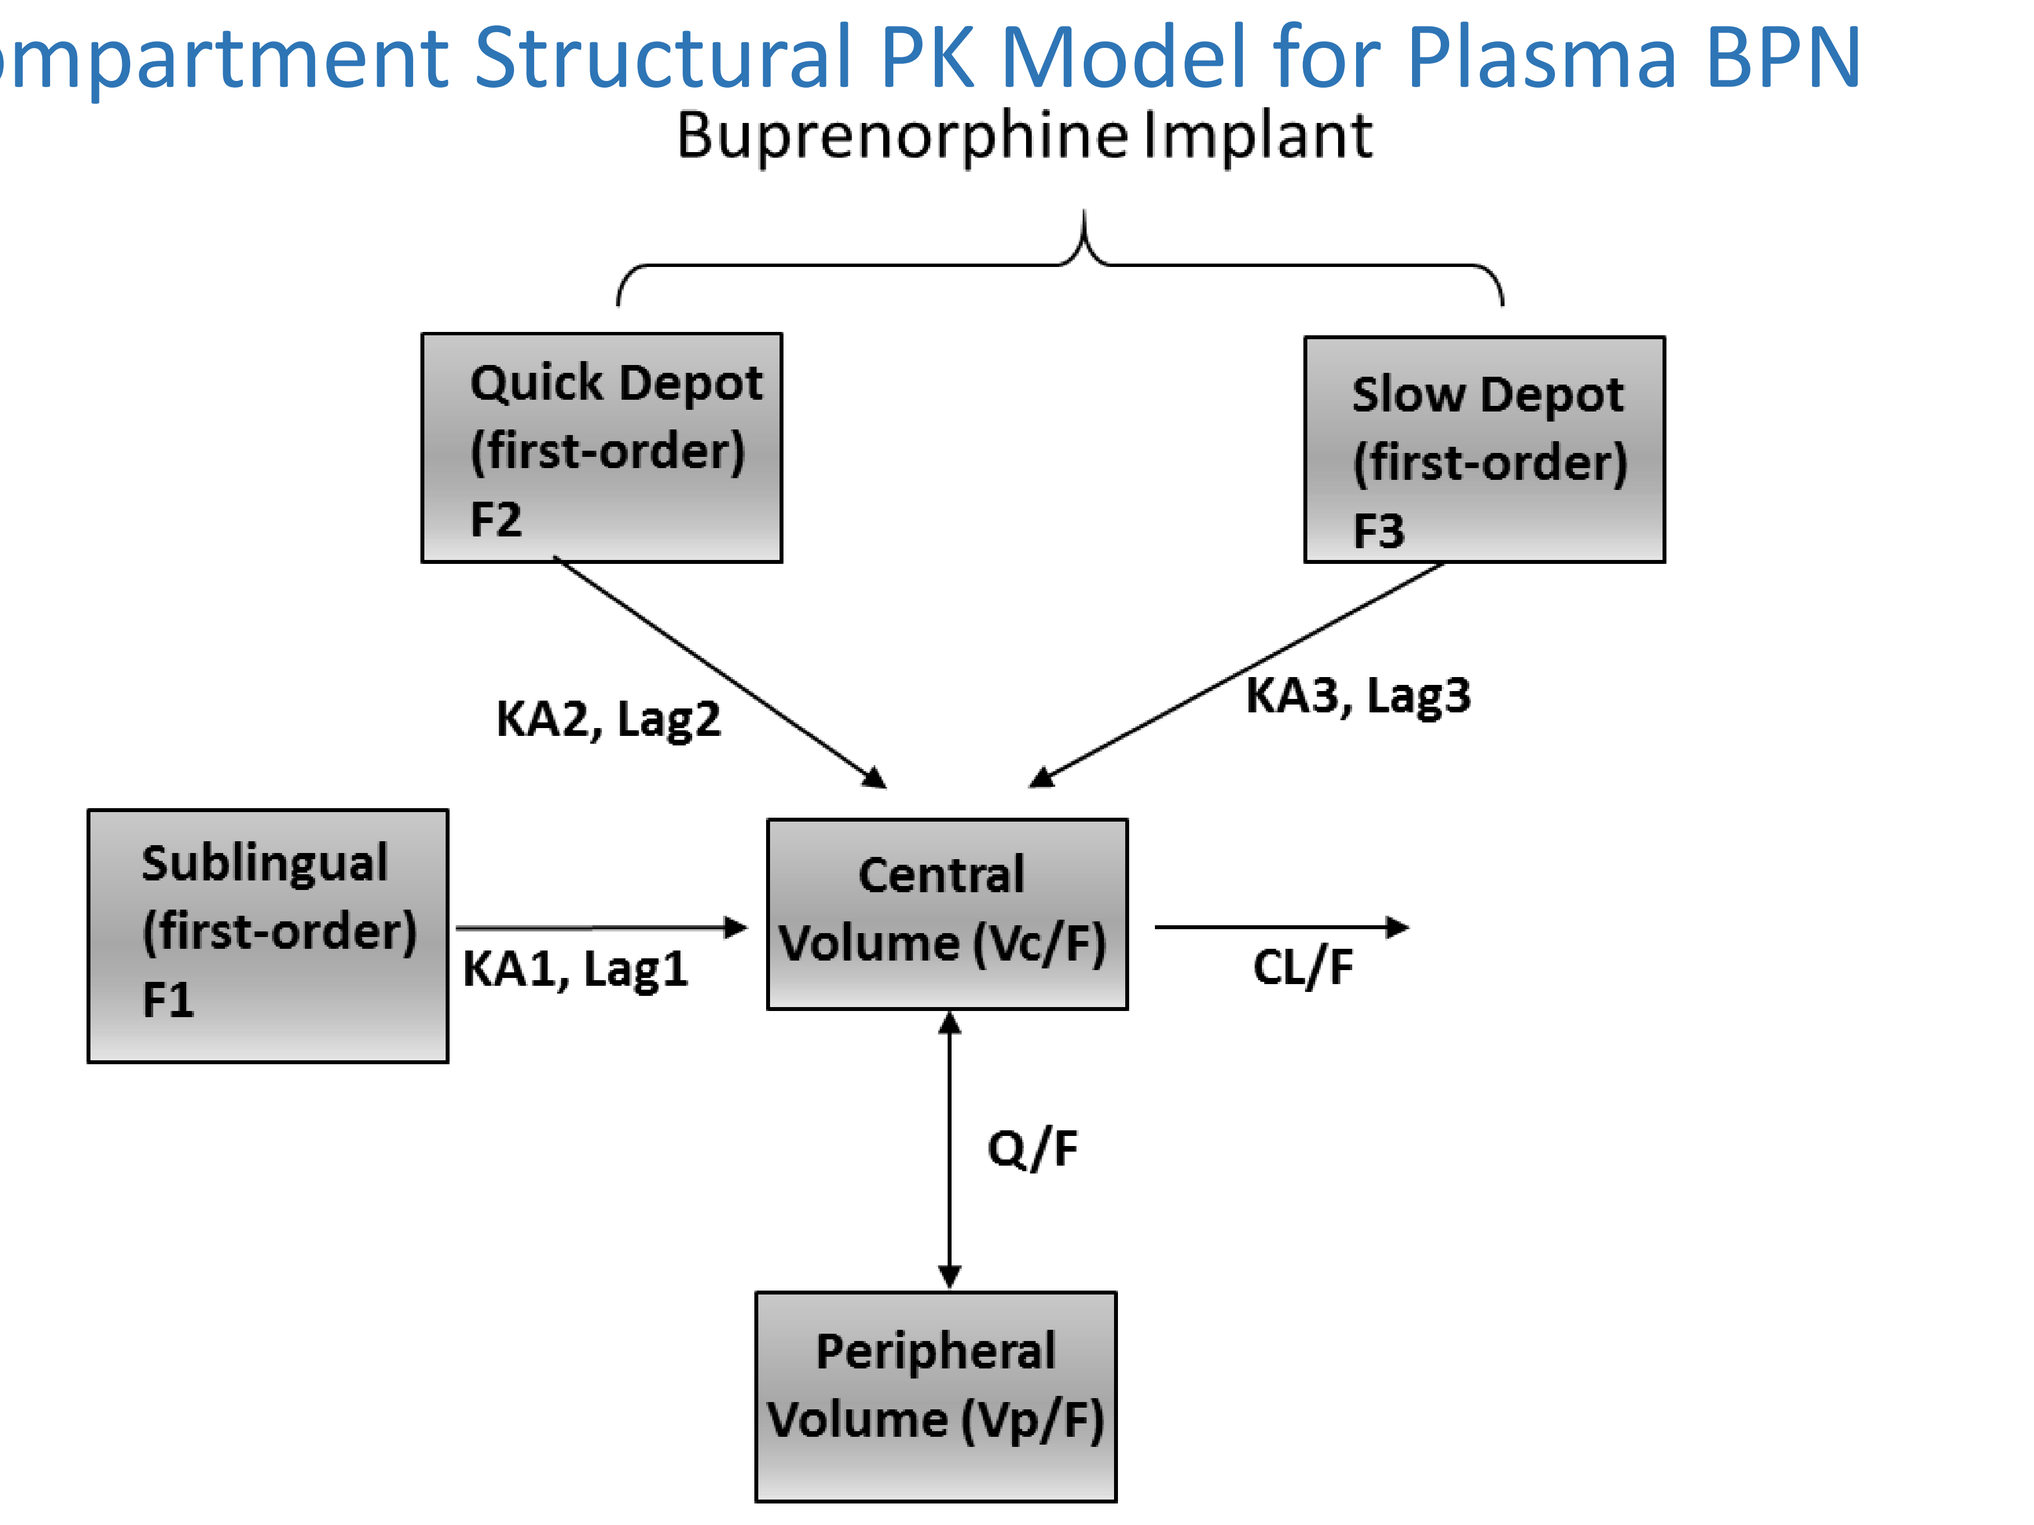
Buprenorphine Implant
Quick Depot (first-order) F2
Slow Depot (first-order) F3
KA2, Lag2
KA3, Lag3
Sublingual (first-order) F1
KA1, Lag1
Central Volume (Vc/F)
CL/F
Q/F
Peripheral Volume (Vp/F)

BPN = buprenorphine, CL/F = apparent clearance; KA1, KA2, KA3= first-order rate constant of absorption of compartments 1, 2 and 3, respectively; F1, F2, F3= relative fraction of absorption for compartments 1, 2 and 3, respectively; Lag1, Lag2, Lag3= lag time of absorption for compartments 1, 2 and 3, respectively; PK= pharmacokinetic; Q/F= apparent inter-compartmental clearance; Vc/F= apparent central volume of distribution; Vp/F= apparent peripheral volume of distribution.

Figure 2. Simulated Rich Concentration-Time Profiles of Plasma BPN in Subjects with Probuphine Implants

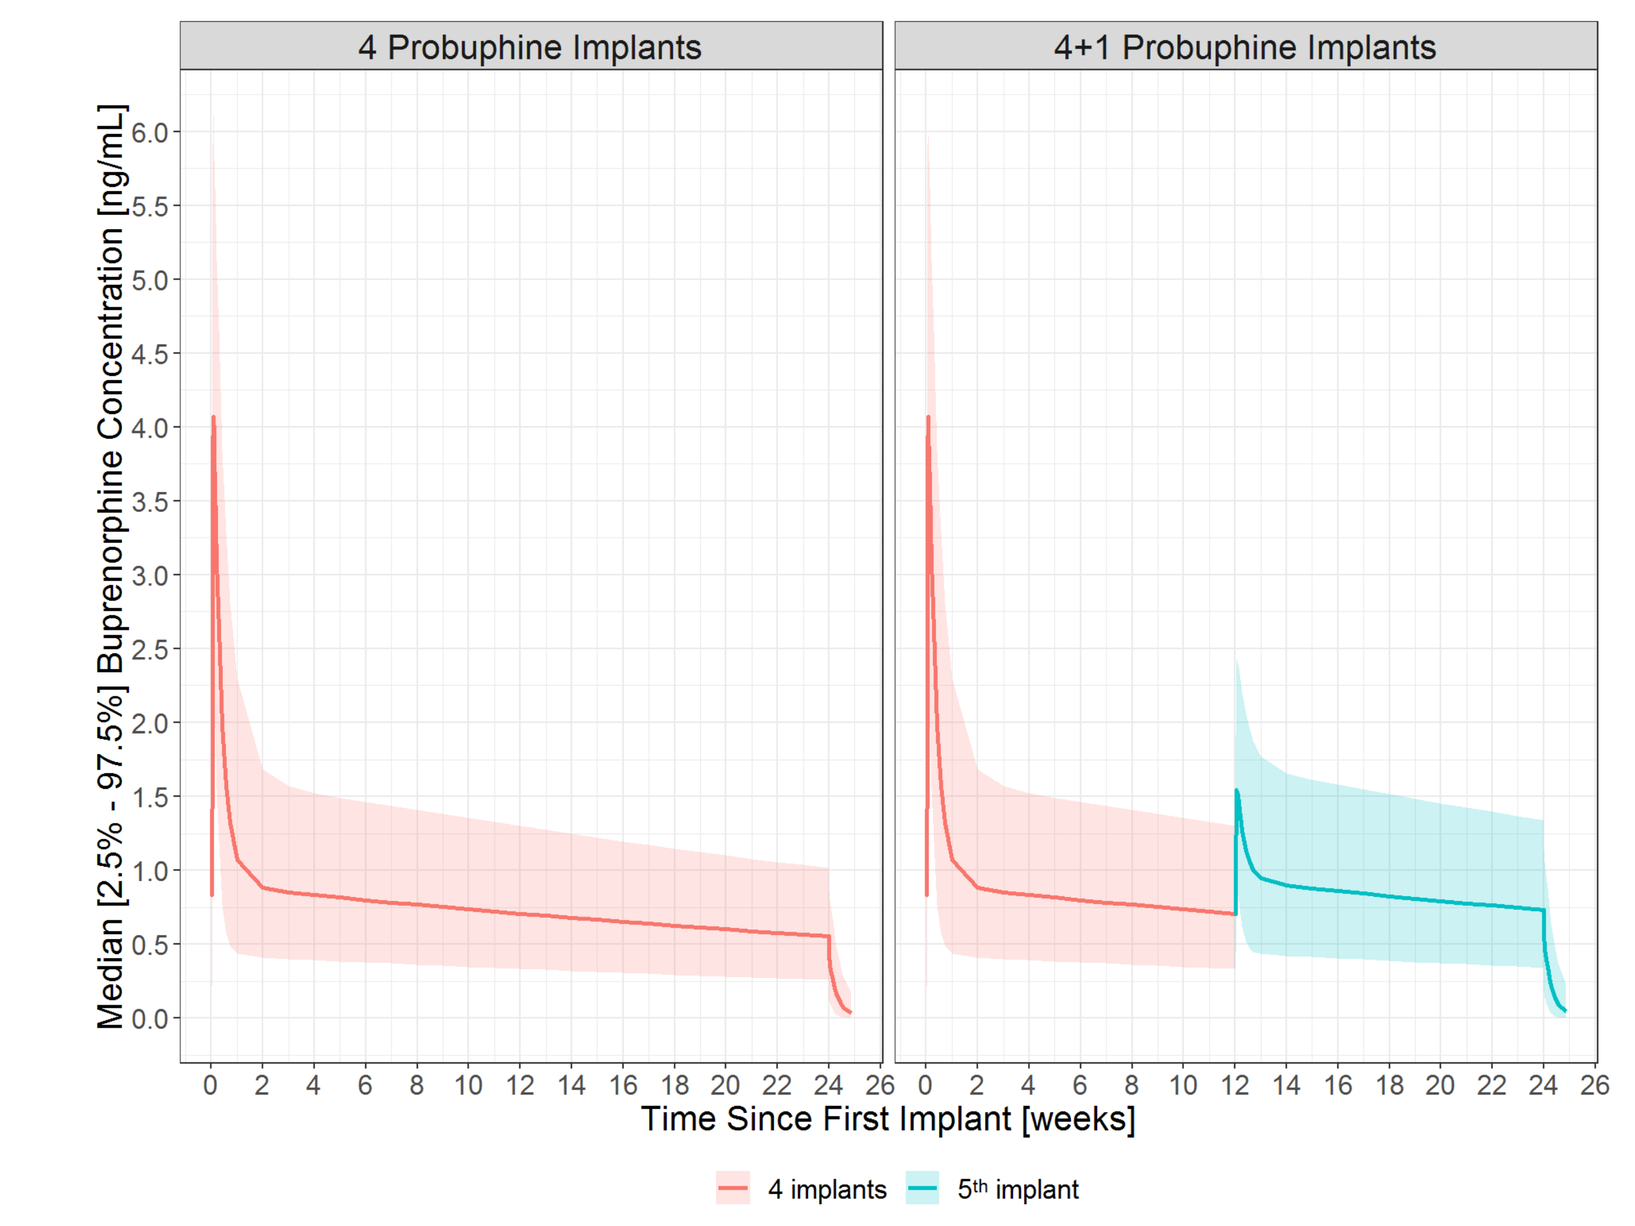
4 Probuphine Implants
4+1 Probuphine Implants
Median [2.5% - 97.5%] Buprenorphine Concentration [ng/mL]
Time Since First Implant [weeks]
4 implants
5th implant

Figure 3. Emax Model for E-R Analysis of Plasma BPN Cavg' and Percentage of Opioid Negative Urines

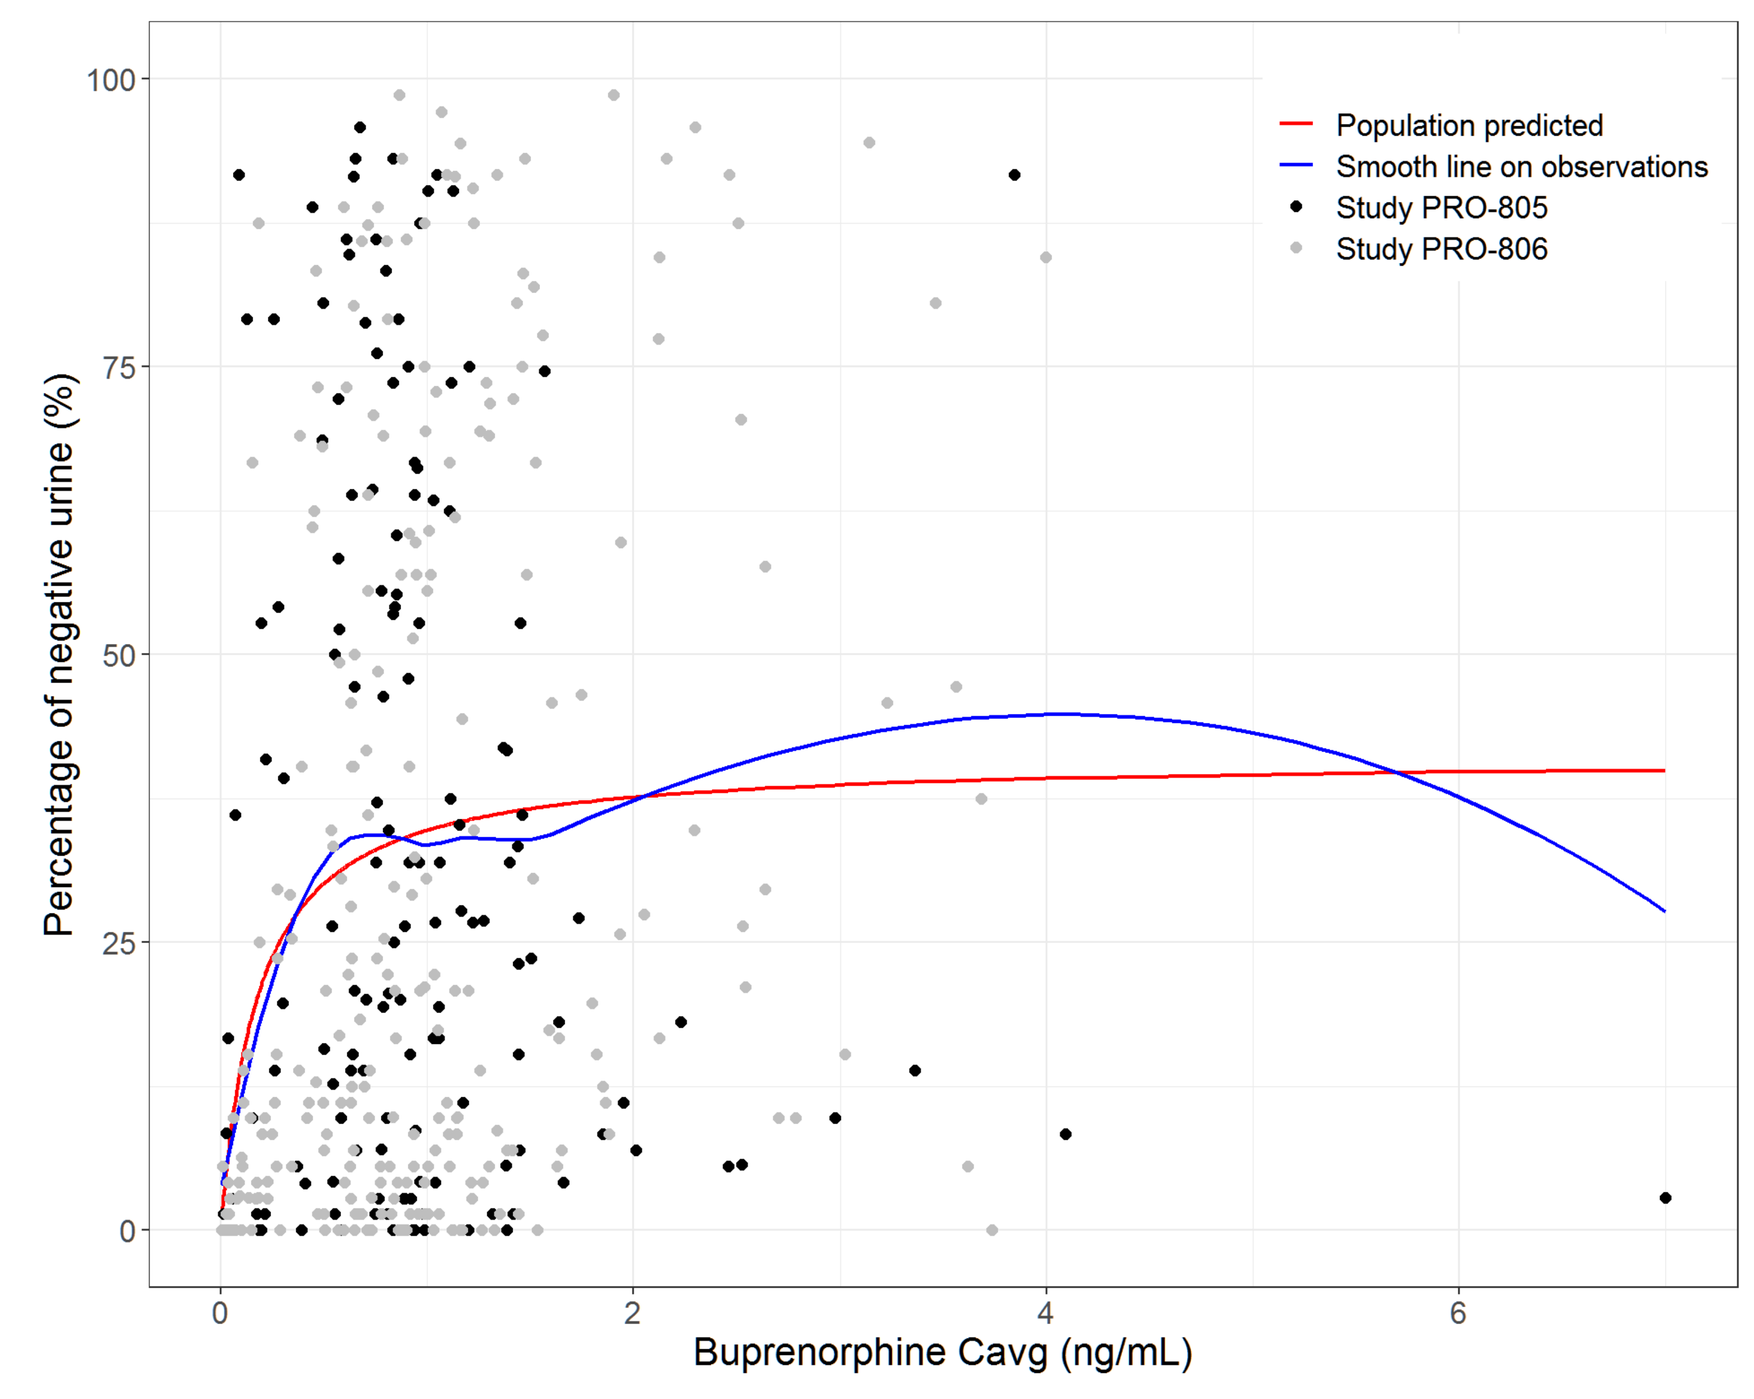
Population predicted
Smooth line on observations
Study PRO-805
Study PRO-806
Percentage of negative urine (%)
Buprenorphine Cavg (ng/mL)

Cavg'= average concentration at Weeks 1-24; Note: Dashed vertical lines represent the limits of the quartiles of Cavg' of plasma BPN.

Figure 4. Logistic Regression Between Responder Percentage with Opioid Negative Urines >30% and Plasma BPN Cavg' for Treatment Weeks 1-24

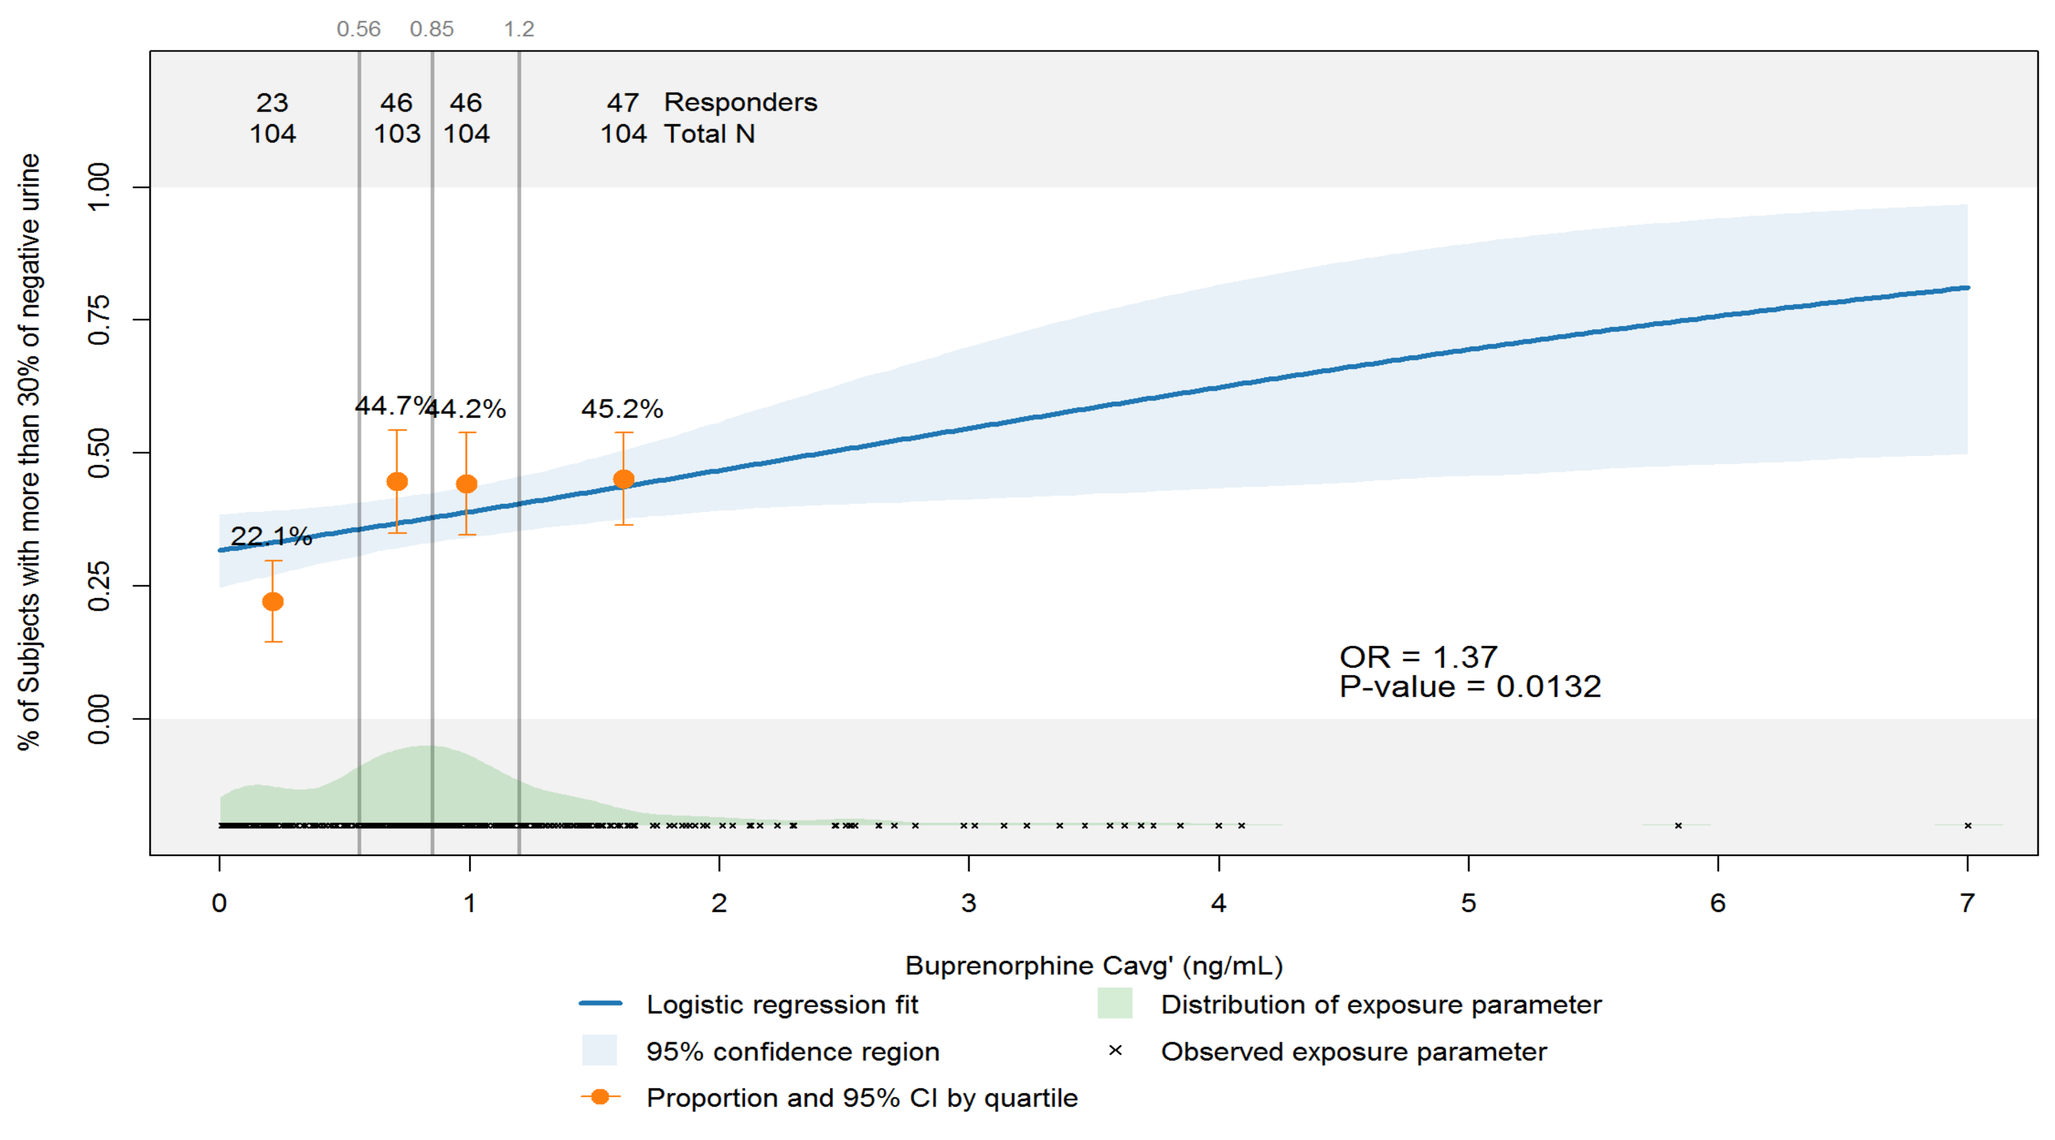
0.56 0.85 1.2
23 104
46 103
46 104
47 104
Responders
Total N
44.7%
44.2%
45.2%
22.1%
OR = 1.37
P-value = 0.0132
% of Subjects with more than 30% of negative urine
Buprenorphine Cavg' (ng/mL)
Logistic regression fit
95% confidence region
Proportion and 95% CI by quartile
Distribution of exposure parameter
Observed exposure parameter