Oral Sulopenem/probenecid for Uncomplicated Urinary Tract Infections (uUTI): Results from the REASSURE Trial

Sailaja Puttagunta, MD[1], Steven I. Aronin, MD[2], Jayanti Gupta, PhD[2], Anita F. Das, PhD[3], Kalpana Gupta, MD[4] and Michael W. Dunne, MD[1,5]
[1]Past employee, Iterum Therapeutics; [2]Iterum Therapeutics, Old Saybrook, CT; [3]AD Stat Consulting, Guerneville, CA; [4]Boston Veterans Affairs Healthcare System and Boston University School of Medicine, Boston, MA;
[5]Current affiliation: Bill & Melinda Gates Medical Research Institute, Cambridge, MA

ABSTRACT

Background
Existing oral antibiotics for treatment of uUTI are not reliably effective due to rising antimicrobial resistant uropathogens. Sulopenem is a broad-spectrum IV/oral penem being developed for treatment of multidrug resistant infections. We conducted a pivotal Phase 3 randomized, double-blind, double-dummy, active controlled trial to evaluate the safety and efficacy of sulopenem/probenecid (SUL) vs amoxicillin/clavulanate (AMC) for treatment of uUTI.

Methods
Adult women with uUTI were randomized to SUL or AMC, both bid for 5 days. The primary objective was to establish noninferiority of SUL to AMC in the mMITT population (patients with ≥10^5 CFU/mL Enterobacterales in baseline urine culture). The primary endpoint was overall success (combined clinical and microbiologic success) at the Test of Cure (TOC) visit. Using a pre-specified procedure to control for multiplicity, mMITT patients with baseline pathogens susceptible to AMC (mMITTS) were tested for noninferiority/superiority and those with nonsusceptible pathogens (mMITTR) were tested for superiority.

Results
Of 2222 women randomized, 990 (44.6%) had ≥10^5 CFU/mL Enterobacterales in baseline urine cultures and were in the mMITT population (Table 1). Of these, 922 had pathogens susceptible to AMC (mMITTS). The sample size in the mMITTR population (N=67) was lower than anticipated and not sufficiently powered to draw any conclusions.

Table. Overall Success at TOC (Day 12)

Primary end point Population	Sulopenem/probenecid n/N (%)	Amoxicillin/clavulanate n/N (%)	Difference (95% Confidence Interval)	p-value
mMITT	318/522 (60.9)	260/468 (55.6)	5.4 (-0.8, 11.5)	0.0437
mMITTS[1]	296/480 (61.7)	243/442 (55.0)	6.7 (0.3, 13.0)	0.0197[2]
mMITTR	22/42 (52.4)	17/25 (68.0)	-15.6 (-37.5, 9.1)	0.8950

[1] primary population for regulatory approval [2]adhoc p value
N=The number of patients in each treatment group in each primary endpoint population; non-inferiority margin =-10%; superiority tested at 2.5% level.

Treatment emergent adverse events (TEAE) occurred more frequently in SUL treated patients (all, 18.9% vs 12.3%; related, 14.0% vs 7.7%) with the most frequent TEAEs being diarrhea, nausea and headache. Premature discontinuation from study drug due to TEAEs was low in both treatment groups (≤1%). No serious adverse events were reported in the SUL group, while 5 (0.5%) occurred in the AMC group.

Conclusions
Sulopenem/probenecid was non-inferior to AMC for the treatment of adult women with uUTI in the mMITT population. In the mMITTS population, SUL demonstrated non-inferiority and based on an ad hoc analysis, demonstrated superiority to AMC. SUL was well tolerated with a safety profile consistent with other β-lactams and has the potential to fill the substantial unmet medical need for an empiric oral antibiotic option for outpatients with uUTI in this era of rising antimicrobial resistance.

INTRODUCTION

Existing oral antibiotics for treatment of uncomplicated UTI (uUTI) are not reliably effective due to rising rates of antimicrobial resistance. Sulopenem is a broad-spectrum penem with IV and oral formulations being developed for treatment of multidrug resistant infections. We conducted a pivotal Phase 3 randomized, double-blind, double-dummy, active controlled trial to evaluate the safety and efficacy of sulopenem etzadroxil/probenecid (SUL) vs amoxicillin/clavulanate (AMC) for treatment of uUTI.

METHODS

Adult women with uUTI were randomized to SUL or AMC, both bid for 5 days (Figure 1). The primary objective was to establish noninferiority of SUL to AMC in the mMITT population (patients with ≥ 10^5 CFU/mL Enterobacterales in baseline urine culture). The primary endpoint was overall success (combined clinical and microbiologic success at the TOC visit). A pre-specified procedure was used to control for multiplicity (Table 1).

Figure 1: Trial Design

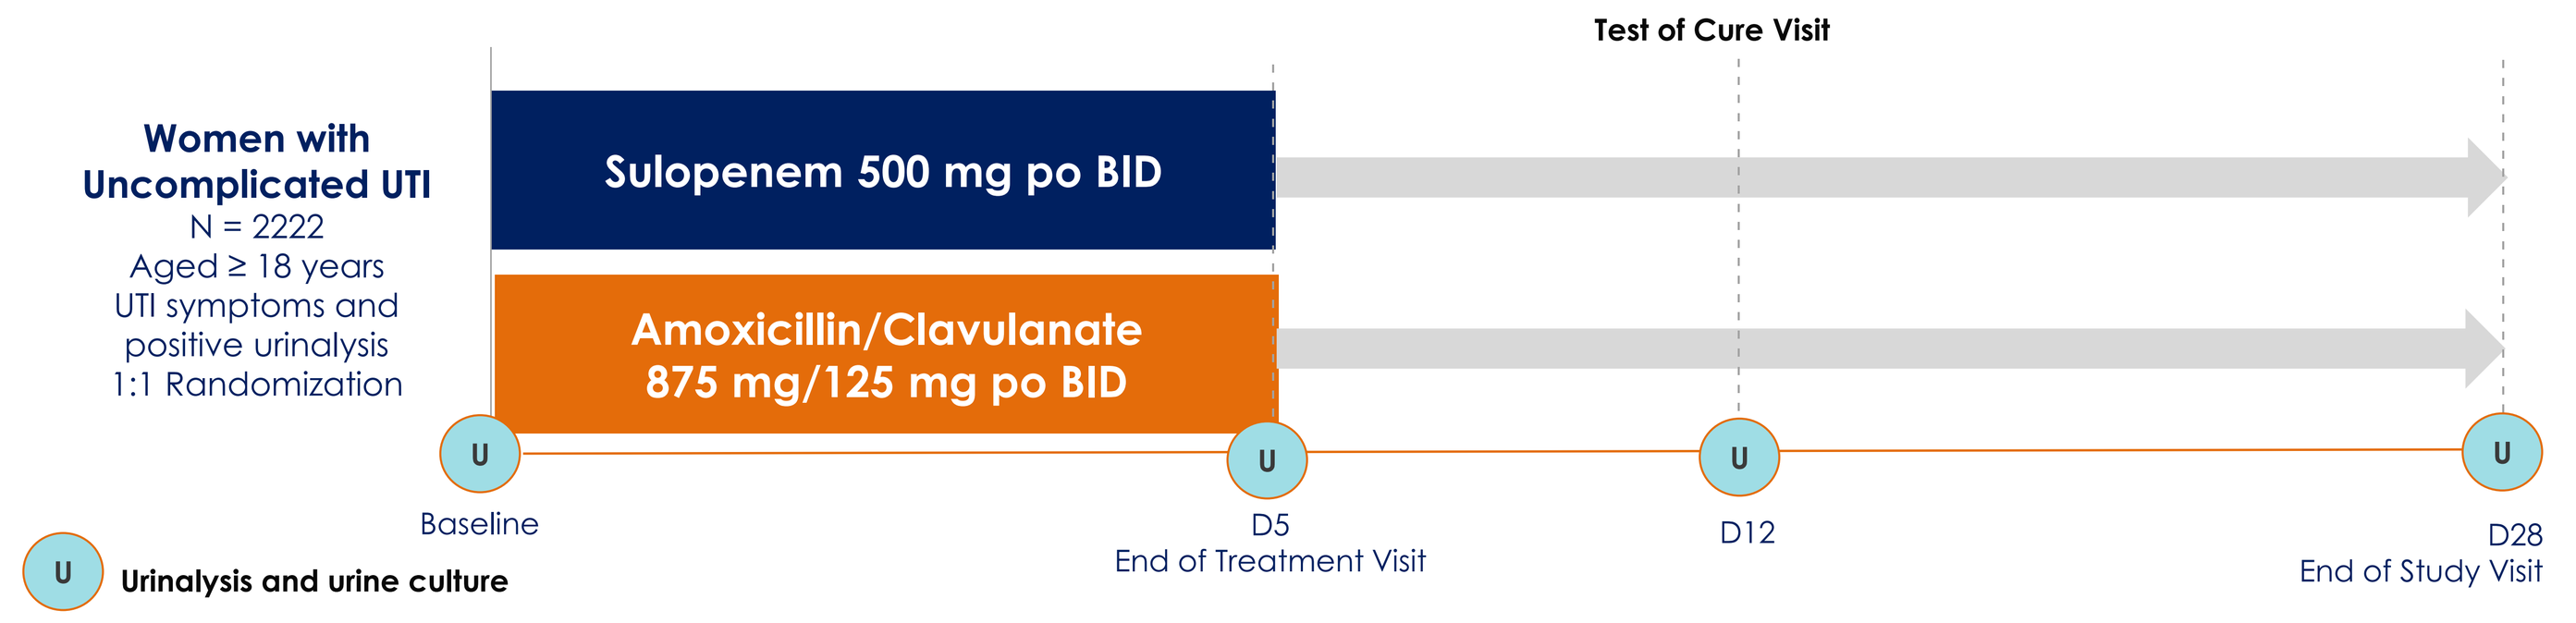
Primary Endpoint
Test of Cure Visit
Women with Uncomplicated UTI
N = 2222
Aged ≥ 18 years
UTI symptoms and positive urinalysis
1:1 Randomization
Sulopenem 500 mg po BID
Amoxicillin/Clavulanate
875 mg/125 mg po BID
U
Baseline
D5
End of Treatment Visit
D12
D28
End of Study Visit
Urinalysis and urine culture

Table 1: Pre-Specified Hierarchical Testing Method of Primary Endpoint

Analysis	Populations	
1st Step	1 **micro-MITT** Non-inferiority of oral sulopenem vs amoxicillin/clavulanate (AMC) in uUTI patients with ≥10^5 CFU/mL of Enterobacterales at baseline	
2nd Step	2 **micro-MITTS** Non-inferiority of oral sulopenem vs AMC in patients with uropathogen susceptible to AMC*	2 **micro-MITTR** Superiority of oral sulopenem vs AMC in patients with uropathogen non-susceptible to AMC

*Primary objective for regulatory approval

RESULTS

Table 2: Study Disposition

	Oral Sulopenem	Amoxicillin/Clavulanate
Intent-to-treat (ITT)	1111	1111
Safety / Modified ITT (MITT) Received study drug	1107	1107
micro-MITT, % (n) Uropathogen ≥ 10^5 CFU/mL	**47.0%** (522)	**42.1%** (468)
micro-MITTS, % (n) Susceptible to amoxicillin/clavulanate	**43.2%** (480)	**39.8%** (442)
micro-MITTR*, % (n) Non-susceptible to amoxicillin/clavulanate	**3.8%** (42)	**2.3%** (25)

*The sample size in the mMITTR population (N=67) was lower than anticipated and not sufficiently powered to draw any conclusions

Table 3: Baseline Demographics, micro-MITT Population

	Oral Sulopenem N = 522	Amoxicillin/Clavulanate N = 468
Age: mean (SD) (years)	**50.3** (17.3)	**48.6** (17.2)
White	**80.3%**	**79.1%**
Black	**16.1%**	**17.9%**
Hispanic / Latinx	**63.8%**	**63.2%**
US	**100%**	**100%**
Diabetes mellitus	**16.5%**	**14.5%**
BMI, median (kg/m^2)	**28.1**	**27.9**
Creatinine clearance, median (mL/min)	**83.1**	**83.7**

Table 4: Baseline Uropathogens, micro-MITTS Population

Pathogen % (n)	Oral Sulopenem % (n) N=480	Amoxicillin/Clavulanate % (n) N=442	Total % (n) N=922
# of patients ≥ 1 study uropathogen at baseline	480 (100.0)	442 (100.0)	922 (100.0)
Escherichia coli	**83.3%** (400)	**84.6%** (374)	**83.9%** (774)
Klebsiella pneumoniae	**11.9%** (57)	**11.3%** (50)	**11.6%** (107)
Proteus mirabilis	**2.7%** (13)	**2.9%** (13)	**2.8%** (26)
Klebsiella variicola	**1%** (5)	**0.2%** (1)	**0.7%** (6)
Citrobacter koseri	**0.6%** (3)	**0.5%** (2)	**0.5%** (5)
Klebsiella oxytoca	**0%** (0)	**0.5%** (2)	**0.2%** (2)
Klebsiella spp	**0.2%** (1)	**0.2%** (1)	**0.2%** (2)
Providencia stuartii	**0.2%** (1)	**0.2%** (1)	**0.2%** (2)
Citrobacter freundii	**0.2%** (1)	**0%** (0)	**0.1%** (1)
Enterobacter hormaechei	**0.2%** (1)	**0%** (0)	**0.1%** (1)
Escherichia spp	**0.2%** (1)	**0%** (0)	**0.1%** (1)
Pantoea spp	**0.2%** (1)	**0%** (0)	**0.1%** (1)

Table 5: Primary and Key Secondary Endpoints, micro-MITTS Population

	Oral Sulopenem N = 480	Amoxicillin/ Clavulanate N = 442	Difference (95% CI)
Overall Success (Day 12)	61.7%	55.0%	**6.7%** (0.3, 13.0)
Clinical Success (Day 12)	77.3%	76.7%	**0.6** (-4.8, 6.1)
Microbiologic Success (Day 12)	75.2%	66.7%	**8.5** (2.6, 14.3)

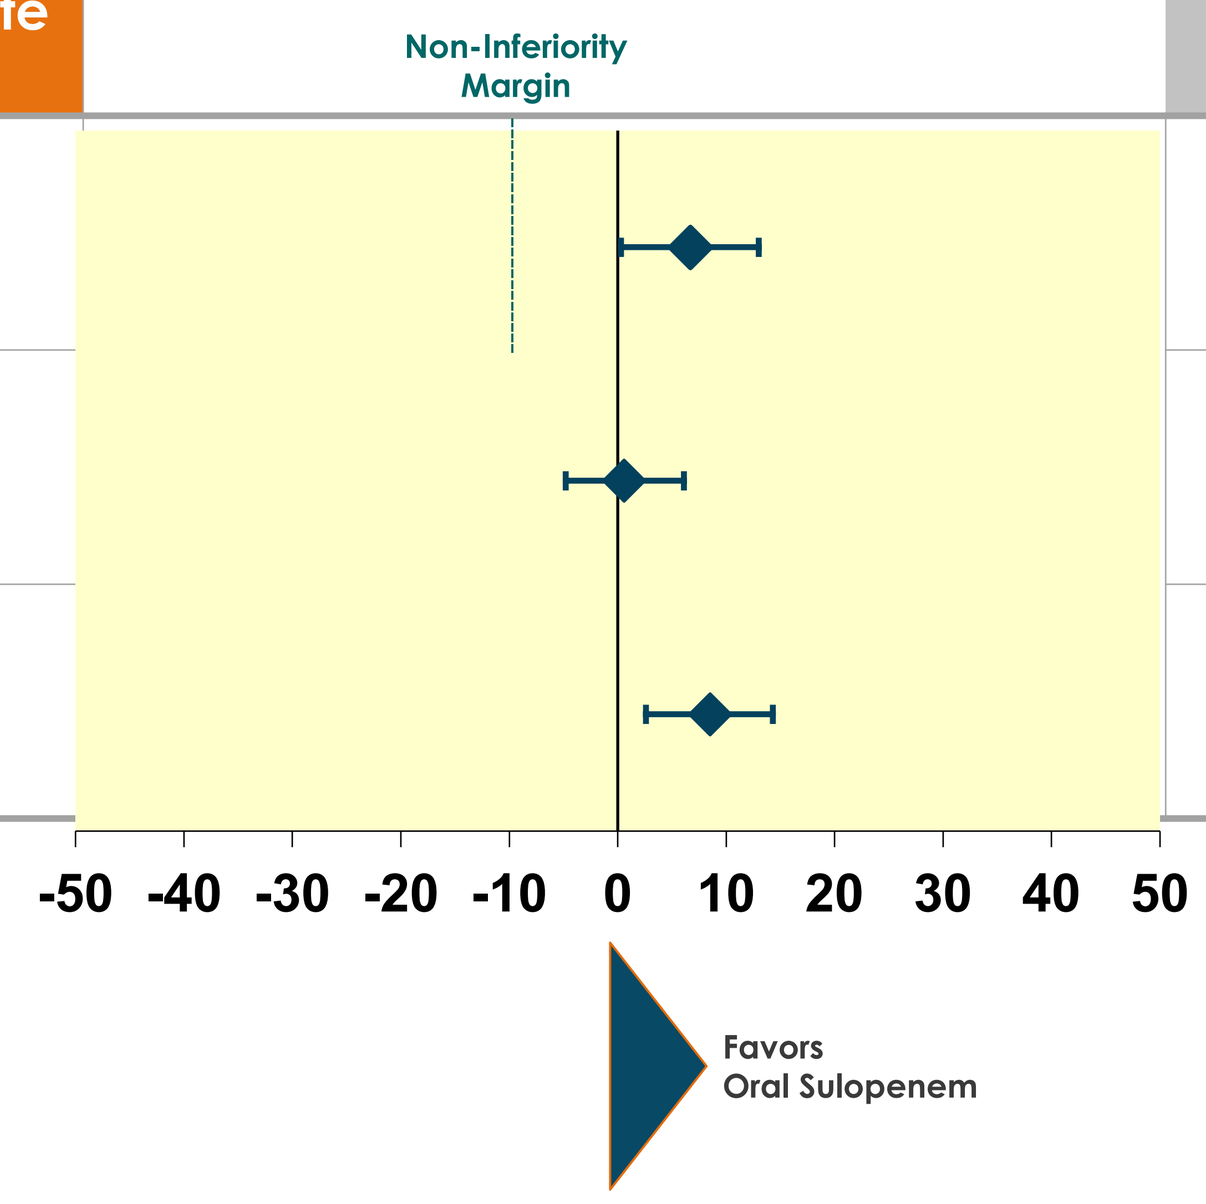
Non-Inferiority Margin
-50 -40 -30 -20 -10 0 10 20 30 40 50
Favors Oral Sulopenem

Table 6: Reasons for Overall Nonresponse at TOC, micro-MITTS Population

Reasons for Failure at TOC, % (n)	Oral Sulopenem N = 480	Amoxicillin/Clavulanate N = 442
Persistent or new uUTI symptoms only	**13.1%** (63)	**10.6%** (47)
Microbiologic failure only (ASB)	**14.6%** (70)	**20.6%** (91)
Both uUTI symptoms and microbiologic failure	**5.4%** (26)	**7.9%** (35)
Non-study antibacterial therapy for uUTI	**1.7%** (8)	**0.9%** (4)

Table 7: Oral Sulopenem versus Amoxicillin/Clavulanate by Visit, micro-MITTS Population

Overall Success	Oral Sulopenem N = 480	Amoxicillin/ Clavulanate N = 442	Difference (95% CI)
End of Treatment (Day 5)	52.5%	51.1%	**1.4** (-5.1, 7.8)
Test of Cure (Day 12)	61.7%	55.0%	**6.7** (0.3, 13.0)
Final Visit (Day 28)	61.5%	55.9%	**5.6** (-0.8, 11.9)

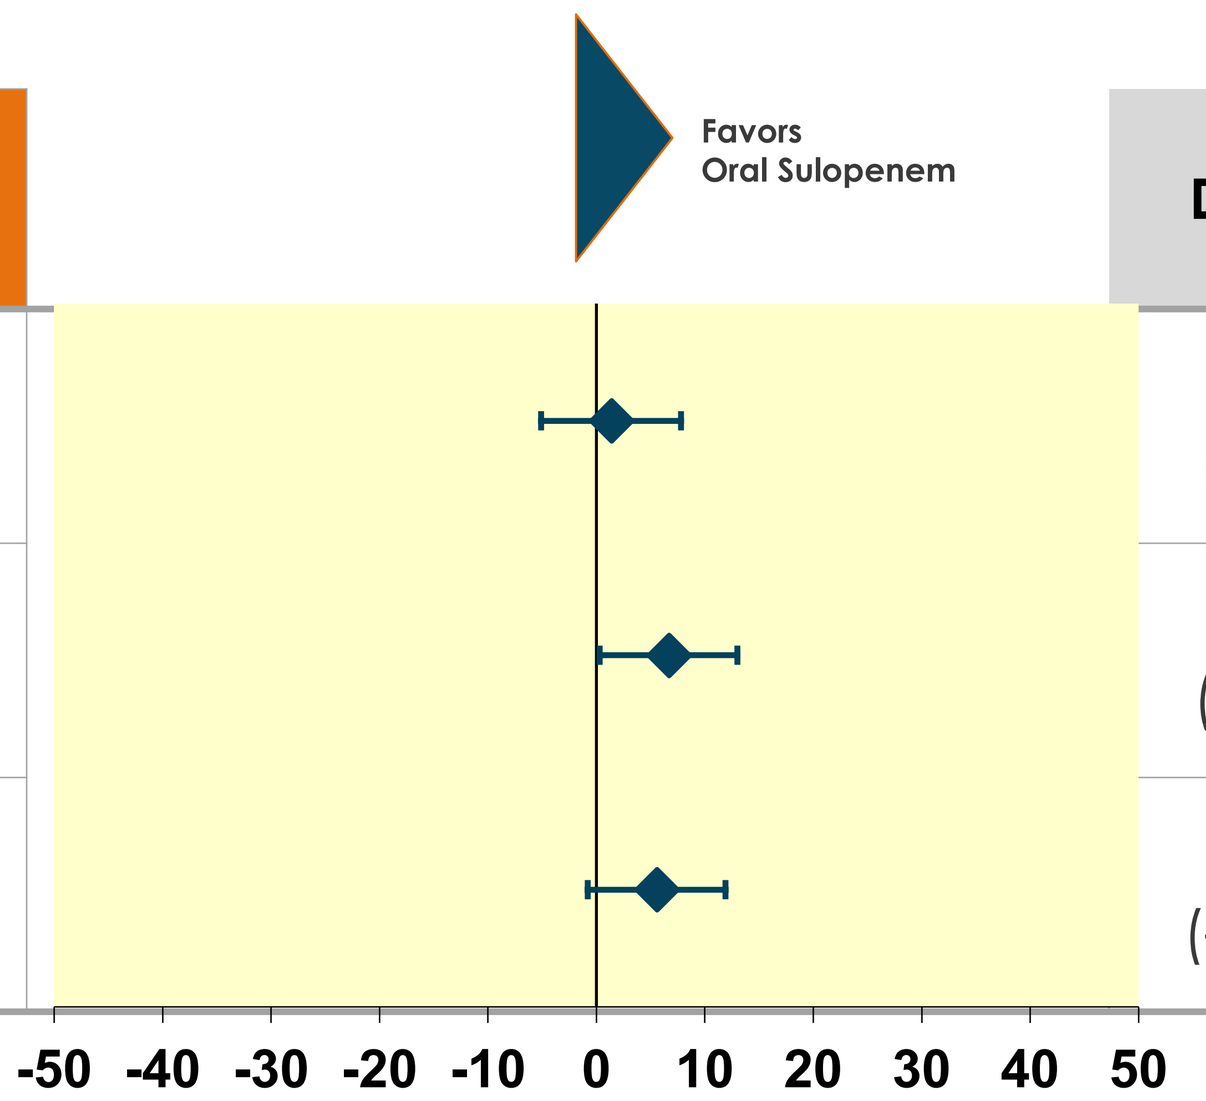
Favors Oral Sulopenem
-50 -40 -30 -20 -10 0 10 20 30 40 50

Figure 2: Sulopenem Treatment Does Not Select for Penem Resistant Organisms, micro-MITT Population

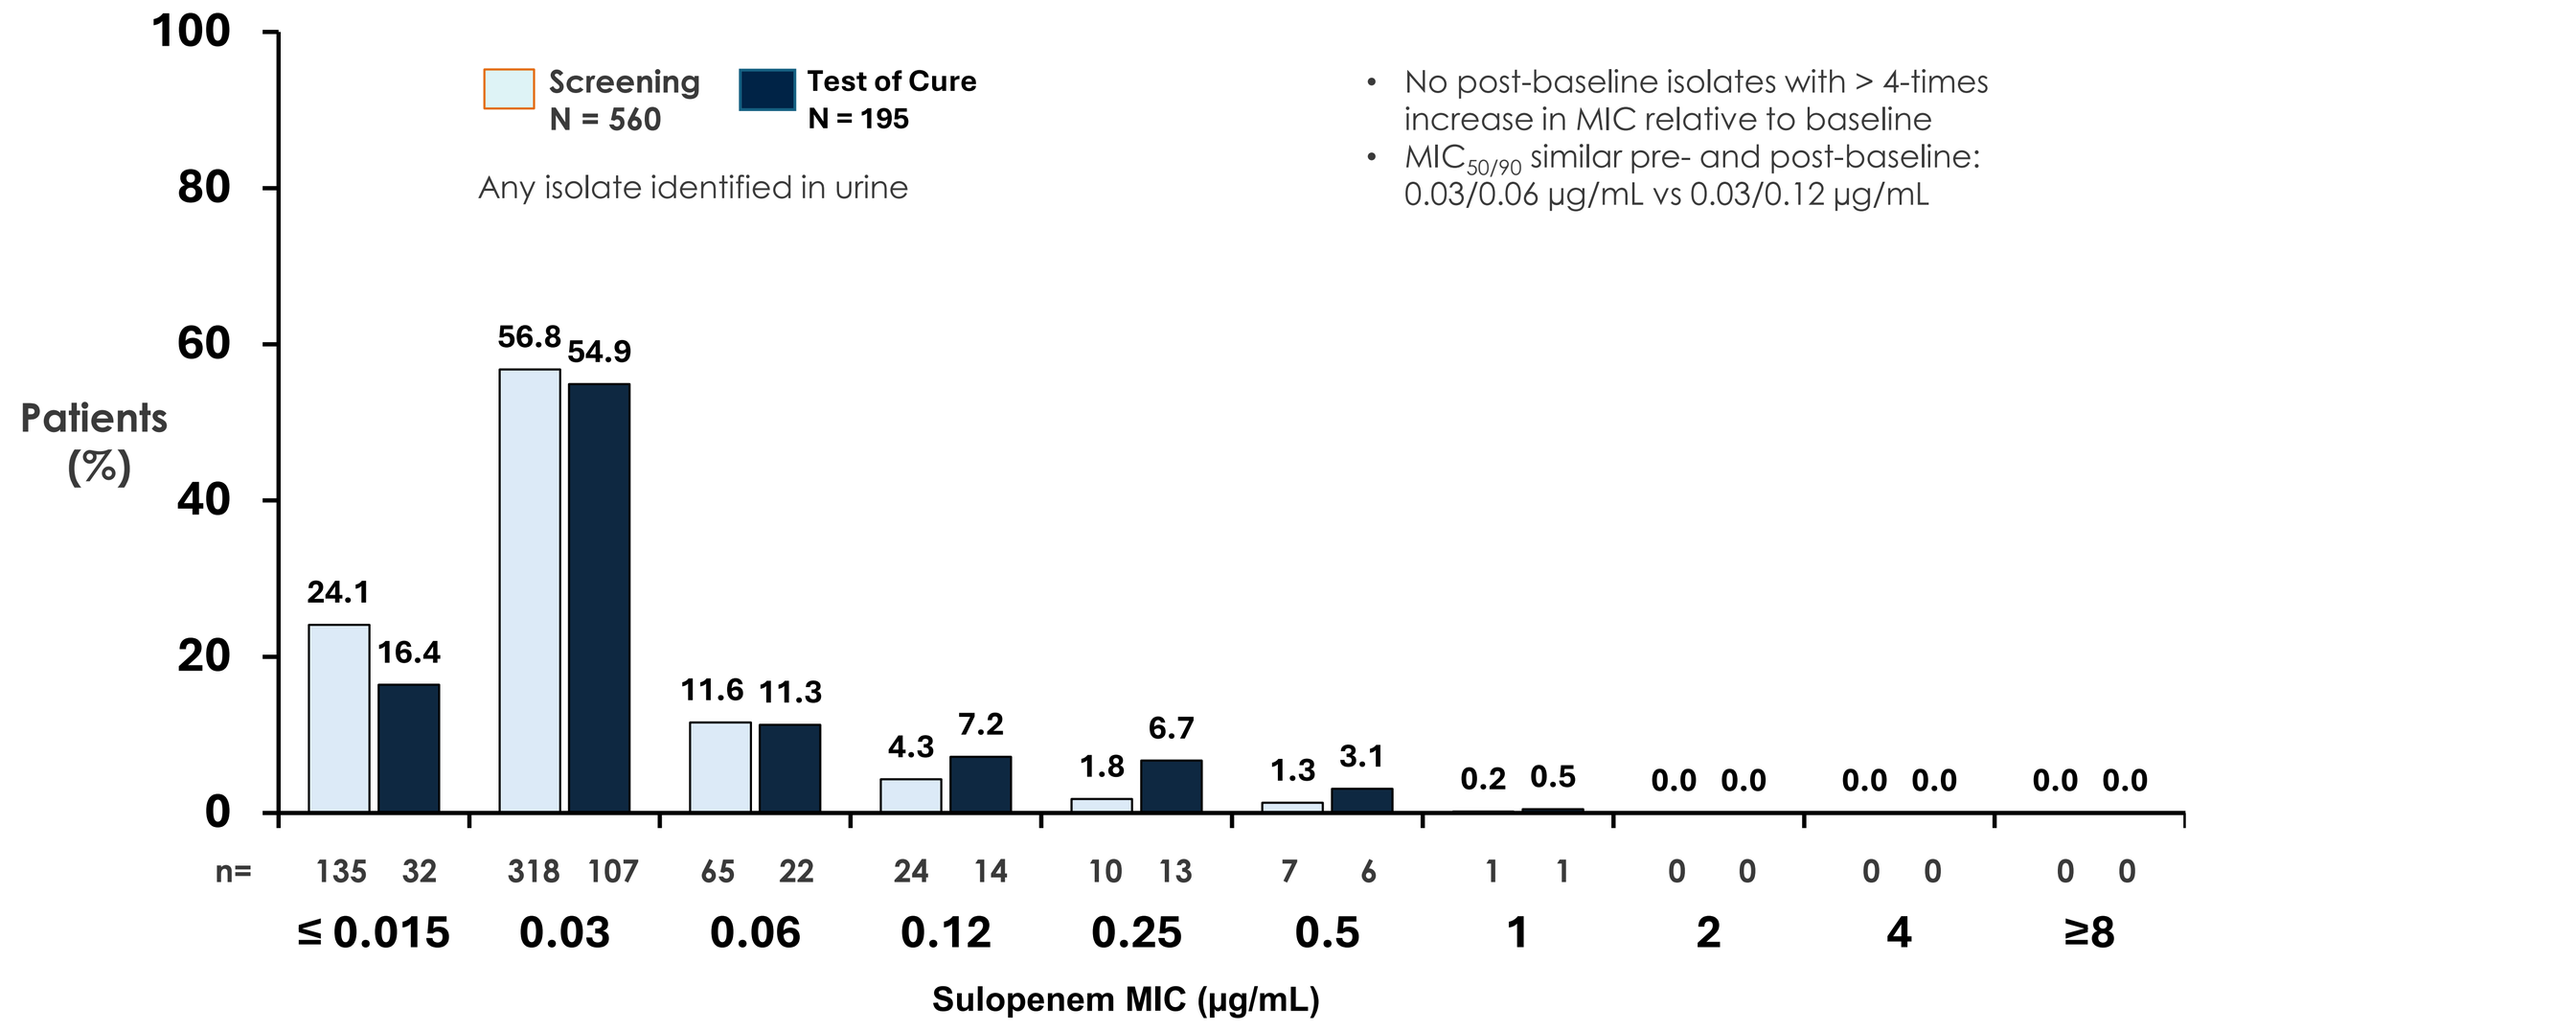
Screening N = 560
Test of Cure N = 195
Any isolate identified in urine
No post-baseline isolates with > 4-times increase in MIC relative to baseline
MIC50/90 similar pre- and post-baseline: 0.03/0.06 µg/mL vs 0.03/0.12 µg/mL
Patients (%)
24.1 16.4
56.8 54.9
11.6 11.3
4.3 7.2
1.8 6.7
1.3 3.1
0.2 0.5
0.0 0.0
0.0 0.0
0.0 0.0
n= 135 32 318 107 65 22 24 14 10 13 7 6 1 1 0 0 0 0 0 0
≤ 0.015 0.03 0.06 0.12 0.25 0.5 1 2 4 ≥8
Sulopenem MIC (µg/mL)

Figure 3: Uropathogens Resistant to Amoxicillin/Clavulanate Identified After Treatment, micro-MITT Population

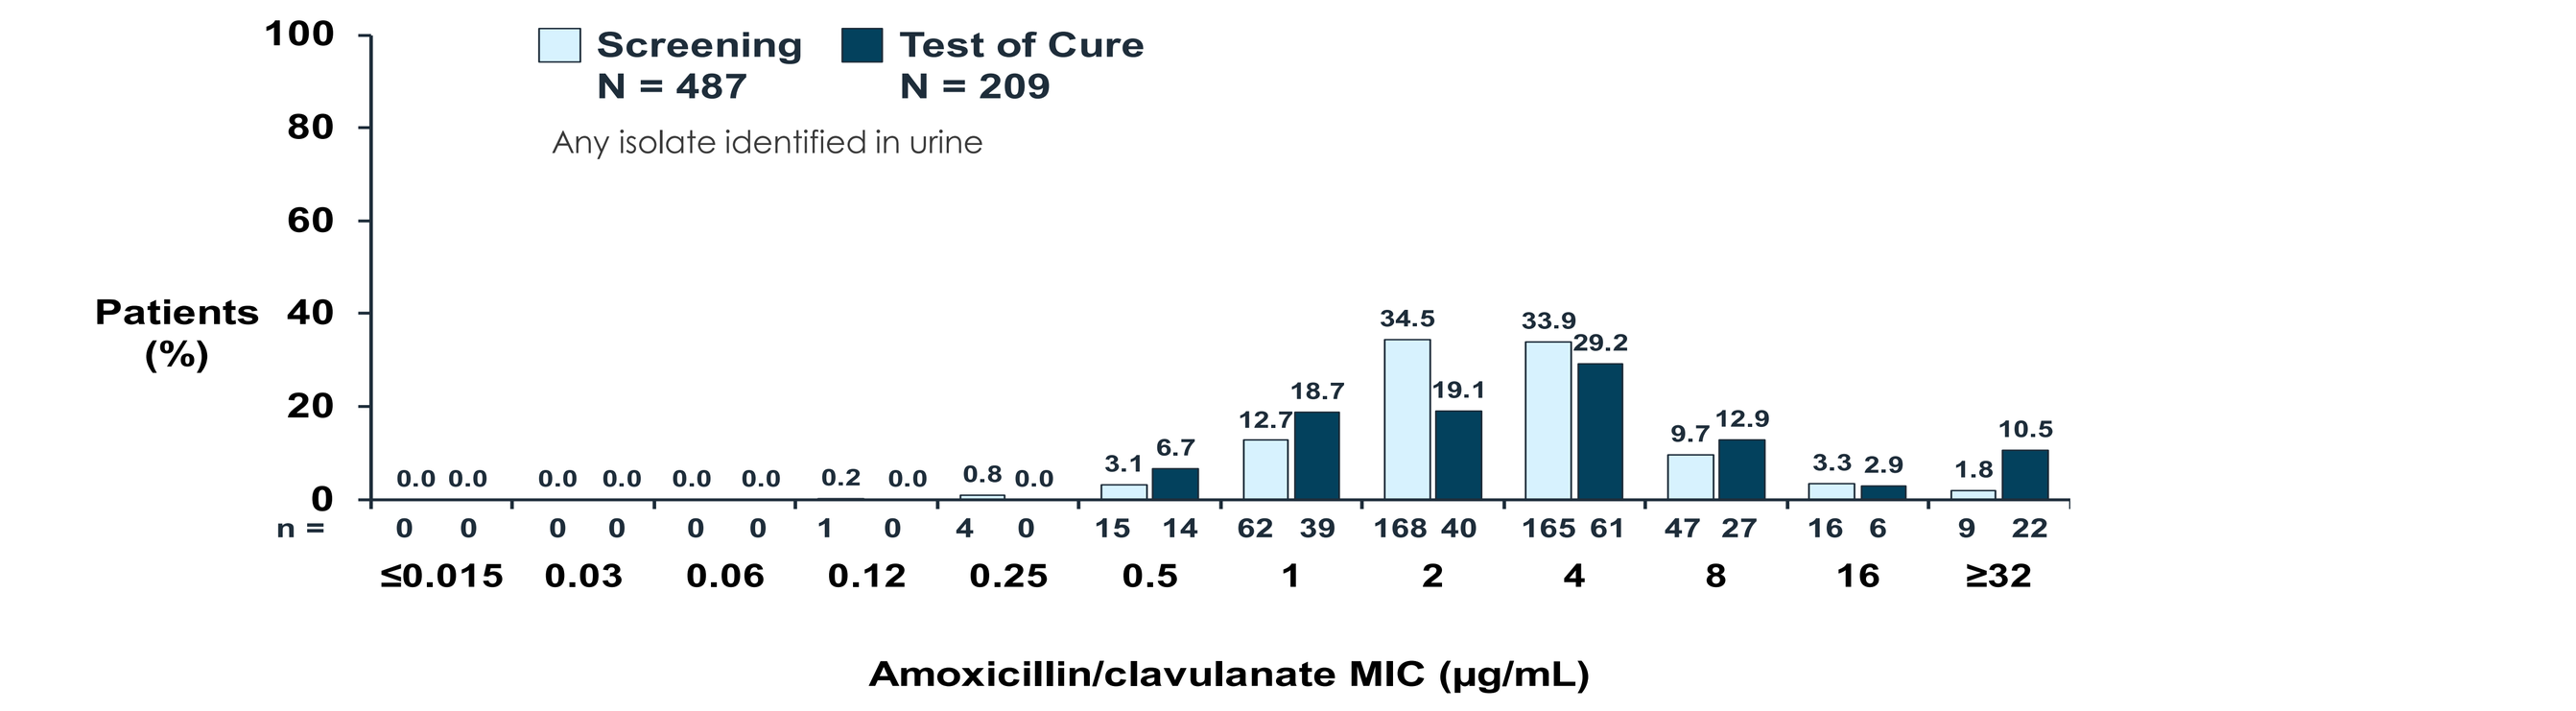
Screening N = 487
Test of Cure N = 209
Any isolate identified in urine
Patients (%)
0.0 0.0 0.0 0.0 0.0 0.0 0.2 0.0 0.8 0.0 3.1 6.7 12.7 18.7 34.5 19.1 33.9 29.2 9.7 12.9 3.3 2.9 1.8 10.5
n = 0 0 0 0 0 0 1 0 4 0 15 14 62 39 168 40 165 61 47 27 16 6 9 22
≤0.015 0.03 0.06 0.12 0.25 0.5 1 2 4 8 16 ≥32
Amoxicillin/clavulanate MIC (µg/mL)

Table 8: Overall Summary of Adverse Events, Safety Population

Parameter % (n)	Oral Sulopenem N =1,107	Amoxicillin /clavulanate N = 1,107
Number of patients who experienced at least one:		
AE	**19.1%** (211)	**12.3%** (136)
TEAE	**18.9%** (209)	**12.3%** (136)
TEAE by maximum severity		
Mild	**71.3%** (149)	**70.6%** (96)
Moderate	**25.8%** (54)	**27.2%** (37)
Severe	**2.4%** (5)	**2.2%** (3)
Drug-related TEAE	**14.0%** (155)	**7.7%** (85)
TEAE leading to discontinuation of study drug	**0.7%** (8)	**0.4%** (4)
TEAE leading to discontinuation from study	**0.4%** (4)	**0.1%** (1)
Serious TEAE (SAE)	**0.0%** (0)	**0.5%** (5)
Drug-related SAE	**0.0%** (0)	**0.0%** (0)
SAE leading to death	**0.0%** (0)	**0.0%** (0)
SAE leading to premature discontinuation of study	**0.0%** (0)	**0.2%** (2)

Table 9: Most Common Adverse Events Occurring in >1% of Patients

Parameter % (n)	Oral Sulopenem N = 1107	Amoxicillin/Clavulanate N = 1107
Diarrhea	**8.1%** (90)	**4.1%** (45)
Nausea	**4.3%** (48)	**2.9%** (32)
Headache	**2.2%** (24)	**1.5%** (17)
Vulvovaginal Mycotic Infection	**1.8%** (20)	**0.4%** (4)
Vomiting	**1.4%** (16)	**0.4%** (4)

CONCLUSIONS

- Oral sulopenem etzadroxil/probenecid (SUL) was non-inferior to amoxicillin/clavulanate (AMC) for the treatment of adult women with uUTI in the mMITT population.
- In the mMITTS population, oral SUL demonstrated non-inferiority and based on the lower limit of the confidence interval being greater than zero, oral SUL demonstrated superiority to AMC.
- No new safety signals were identified beyond those already known for β-lactams.
- Treatment emergent adverse events (TEAE) occurred more frequently in oral SUL treated patients with the most frequent TEAEs being diarrhea, nausea and headache.
- Premature discontinuations from study drug due to TEAEs was low in both treatment groups (≤ 1%).
- No serious adverse events were reported in the oral SUL group, while 5 (0.5%) occurred in the AMC group.
- No *Clostridioides difficile* infections were observed in patients treated with oral SUL.
- Resistance to standard of care antibiotics is increasing at a rapid rate, limiting the options available to safely and effectively treat patients with uUTI.
- Oral SUL meets this unmet medical need and would provide an effective treatment option for patients with uUTI.

REFERENCES

- Puttagunta S, et al. Oral sulopenem versus amoxicillin/clavulanate for the treatment of uncomplicated urinary tract infections (uUTI): A phase 3 randomized trial [Manuscript submitted for publication].
- Dunne MW, et al. Sulopenem or ciprofloxacin for the treatment of uncomplicated urinary tract infections in women: A phase 3, randomized trial. *Clin Infect Dis* 2023;76(1):66-77.
- Dunne MW, et al. A multicenter analysis of trends in resistance in urinary Enterobacterales isolates from ambulatory patients in the United States: 2011-2020. *BMC Infect Dis* 2022;22:194.
- Aronin SI, et al. Regional differences in antibiotic-resistant Enterobacterales urine isolates in the United States: 2018-2020. *Int J Infect Dis* 2022;119:142-145.
- Dunne MW, et al. Impact of empirical antibiotic therapy on outcomes of outpatient urinary tract infection due to nonsusceptible Enterobacterales. *Microbiol Spectrum* 2022;10(1):e02359-21.

Copyright © 2024 Iterum Therapeutics, Old Saybrook, CT, United States.

ITERUM Therapeutics