M146

TRV045, a novel, selective SIP1 receptor modulator is efficacious in acute, chronic and prevention modes in mouse models of chronic neuropathy, without causing lymphopenia

Ruihua Chen, PhD[1], Mark A. Demitrack, MD[1], Torin Honaker[3], Matthew Harding, PhD[2], Dana E. Selley, PhD[3], and M. Imad Damaj, PhD[3,4]

[1]Trevena, Inc., Chesterbrook, PA; [2]Pharmacology Consultant, Meredith, NH; [3]Dept of Pharmacology and Toxicology, [4]Translational Research Initiative for Pain and Neuropathy, Virginia Commonwealth University, Richmond, VA

BACKGROUND

Chemotherapy-induced peripheral neuropathy (CIPN) occurs in up to 70% of oncology patients undergoing chemotherapy. There is an unmet need for prophylactic and symptomatic treatment of CIPN. Previous studies have suggested a beneficial role for non-selective sphingosine-1-phosphate receptor (S1PR) modulators in treating neuropathic pain. However, at doses which produce analgesia, these compounds also affect lymphocyte trafficking, resulting in profound reductions in circulating peripheral lymphocytes. This limits their utility for use in clinical conditions, like CIPN, where immunosuppression would not be desirable.

OBJECTIVE

Evaluate TRV045, a selective S1PR subtype-1 specific modulator that does not cause lymphopenia, following single or repeat oral dosing in a mouse model of CIPN induced by paclitaxel.

METHODS

CIPN Test Model: Paclitaxel (8 mg/kg) or vehicle was administered intraperitoneally (i.p.) once per day on Days 1, 3, 5 and 7. Baseline behaviors were measured on Day 0 and 14. The complete testing methods are described briefly here, the full methods are available elsewhere (Caillaud, 2021).

Mechanical withdrawal thresholds were measured by applying von Frey filaments in incremental force from 0.07 grams to 3.6 grams to the plantar surface of each hind paw with the intent to elicit an appropriate response (paw withdrawn, licking or shaking). Stimulation of the same filament was applied 3x at 10 second intervals. The results are presented as the average of the data from both paws. **Cold hypersensitivity** was assessed via acetone test. 20 µL of acetone (Sigma-Aldrich, MO, USA) was applied onto the plantar surface of each hind paw via pipette. Time spent licking or shaking the hind paw was recorded for 10 seconds. **Voluntary wheel running** test was done by placing mice in polycarbonate wheels (diameter 21.5 cm; width 5 cm) with a steel rod axle containing an electronic sensor recording the number of rotations each 60-minutes. Data are expressed as the number of rotations completed (Figure 5).

Acute Reversal of CIPN Nociception: (Figure 1) Following acquisition of CIPN, on day 15, mice, in a within-subject design, received vehicle or FTY-720 at 0.1 mg/kg (p.o.) and tested 1, 3, 6 and 24 hours after drug administration. On day 17, mice received 0.3 mg/kg of FTY-720, on day 22, 1 mg/kg, and were tested for mechanical and cold hypersensitivity. (Figures 2A and 2B) Following acquisition of CIPN, TRV045, on day 15, male mice received FTY-720 at 1 mg/kg, TRV-045 at different doses (0.1, 0.3, 3 and 10 mg/kg) or its vehicle (10% DMAC + 10% Cremophor EL + 80% [10% HPβCD in water]) via oral gavage (p.o.). Mice were tested for mechanical and cold hypersensitivity at 1, 3 and 6 hr after the drug administration. Female mice study design same as male except without dose of 0.1mg/kg and adding a behavior test time point at 24 hr. For both, mice were tested in the voluntary wheel running assay after the final test of mechanical and cold hypersensitivity.

Chronic Reversal of CIPN Nociception: (Figure 3) Following acquisition of CIPN, on day 15, mice received TRV045 at different doses (1, 3 and 10 mg/kg) or its vehicle (p.o.) repeatedly for 7 days (once per day at 8:00 am). Mice were retested on days 21 (at 1, 3, 6 and 24 hr after the last administration (half of the animals were tested at 24 hr) and 28. In addition, on day 21, mice were tested in the voluntary wheel running assay 6 hr after the last administration of drugs.

CIPN Prevention: (Figure 4) Paclitaxel (8 mg/kg) or vehicle were administered intraperitoneally (i.p.) once per day on days 3, 5 7 and 9. Baseline behaviors were measured on day 10. Mice received TRV045 at different doses (0.3, 3. 10 and 100 mg/kg) or its vehicle daily from day 1 through day 14. Mice were tested for mechanical and cold hypersensitivity on day 15 (24 hr after last dose) and day 22 (168 hr after last dose). Mice were also tested in the voluntary wheel running assay after the hypersensitivity test on day 15.

All data are expressed as mean ± SEM (Standard Error of the Mean). Normality and equal variance were verified by Shapiro-Wilk or Brown-Forsythe tests. Mechanical and cold data were compared using 2-way analysis of variance (ANOVA) with repeated measures, followed by post-hoc Tukey's comparison. Wheel running data were analyzed using a 1-way ANOVA followed by post-hoc Dunnett comparison. Analyses were performed with GraphPad (GraphPad Software 9.3, Inc., La Jolla, CA, USA). Significance was assumed at P <0.05.

RESULTS

Figure 1. Acute Reversal of CIPN: Effect of FTY720 (males)

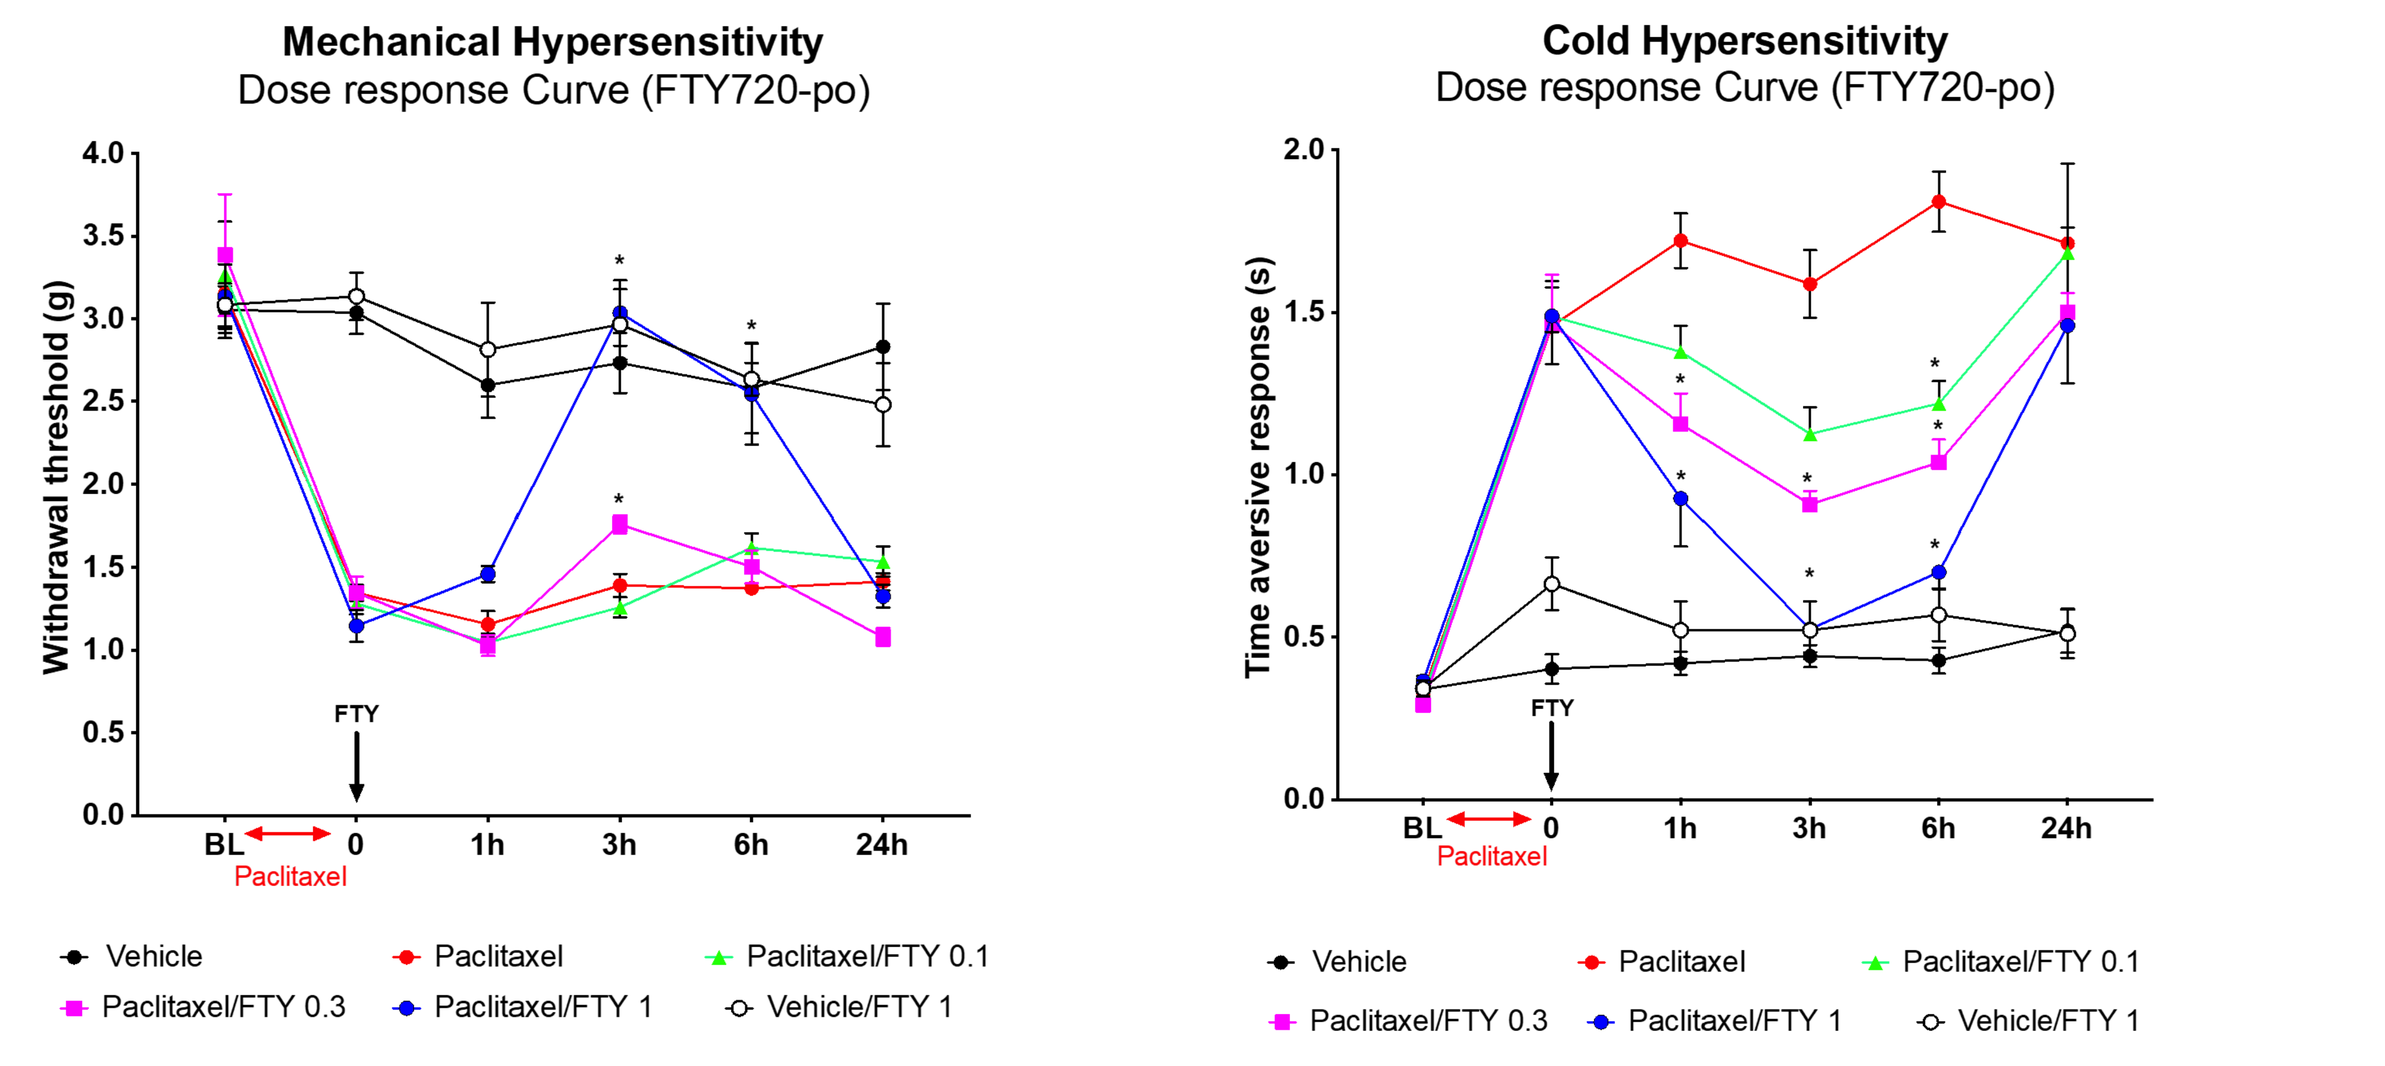
Mechanical Hypersensitivity
Dose response Curve (FTY720-po)
Withdrawal threshold (g)
FTY
Paclitaxel
Cold Hypersensitivity
Dose response Curve (FTY720-po)
Time aversive response (s)
Vehicle
Paclitaxel
Paclitaxel/FTY 0.1
Paclitaxel/FTY 0.3
Paclitaxel/FTY 1
Vehicle/FTY 1

NOTES: Each point represents the mean ± SEM of 8 mice per group. * Denotes significantly different from paclitaxel group.

Figure 2A. Acute Reversal of CIPN: Effect of TRV045 and FTY720 (males)

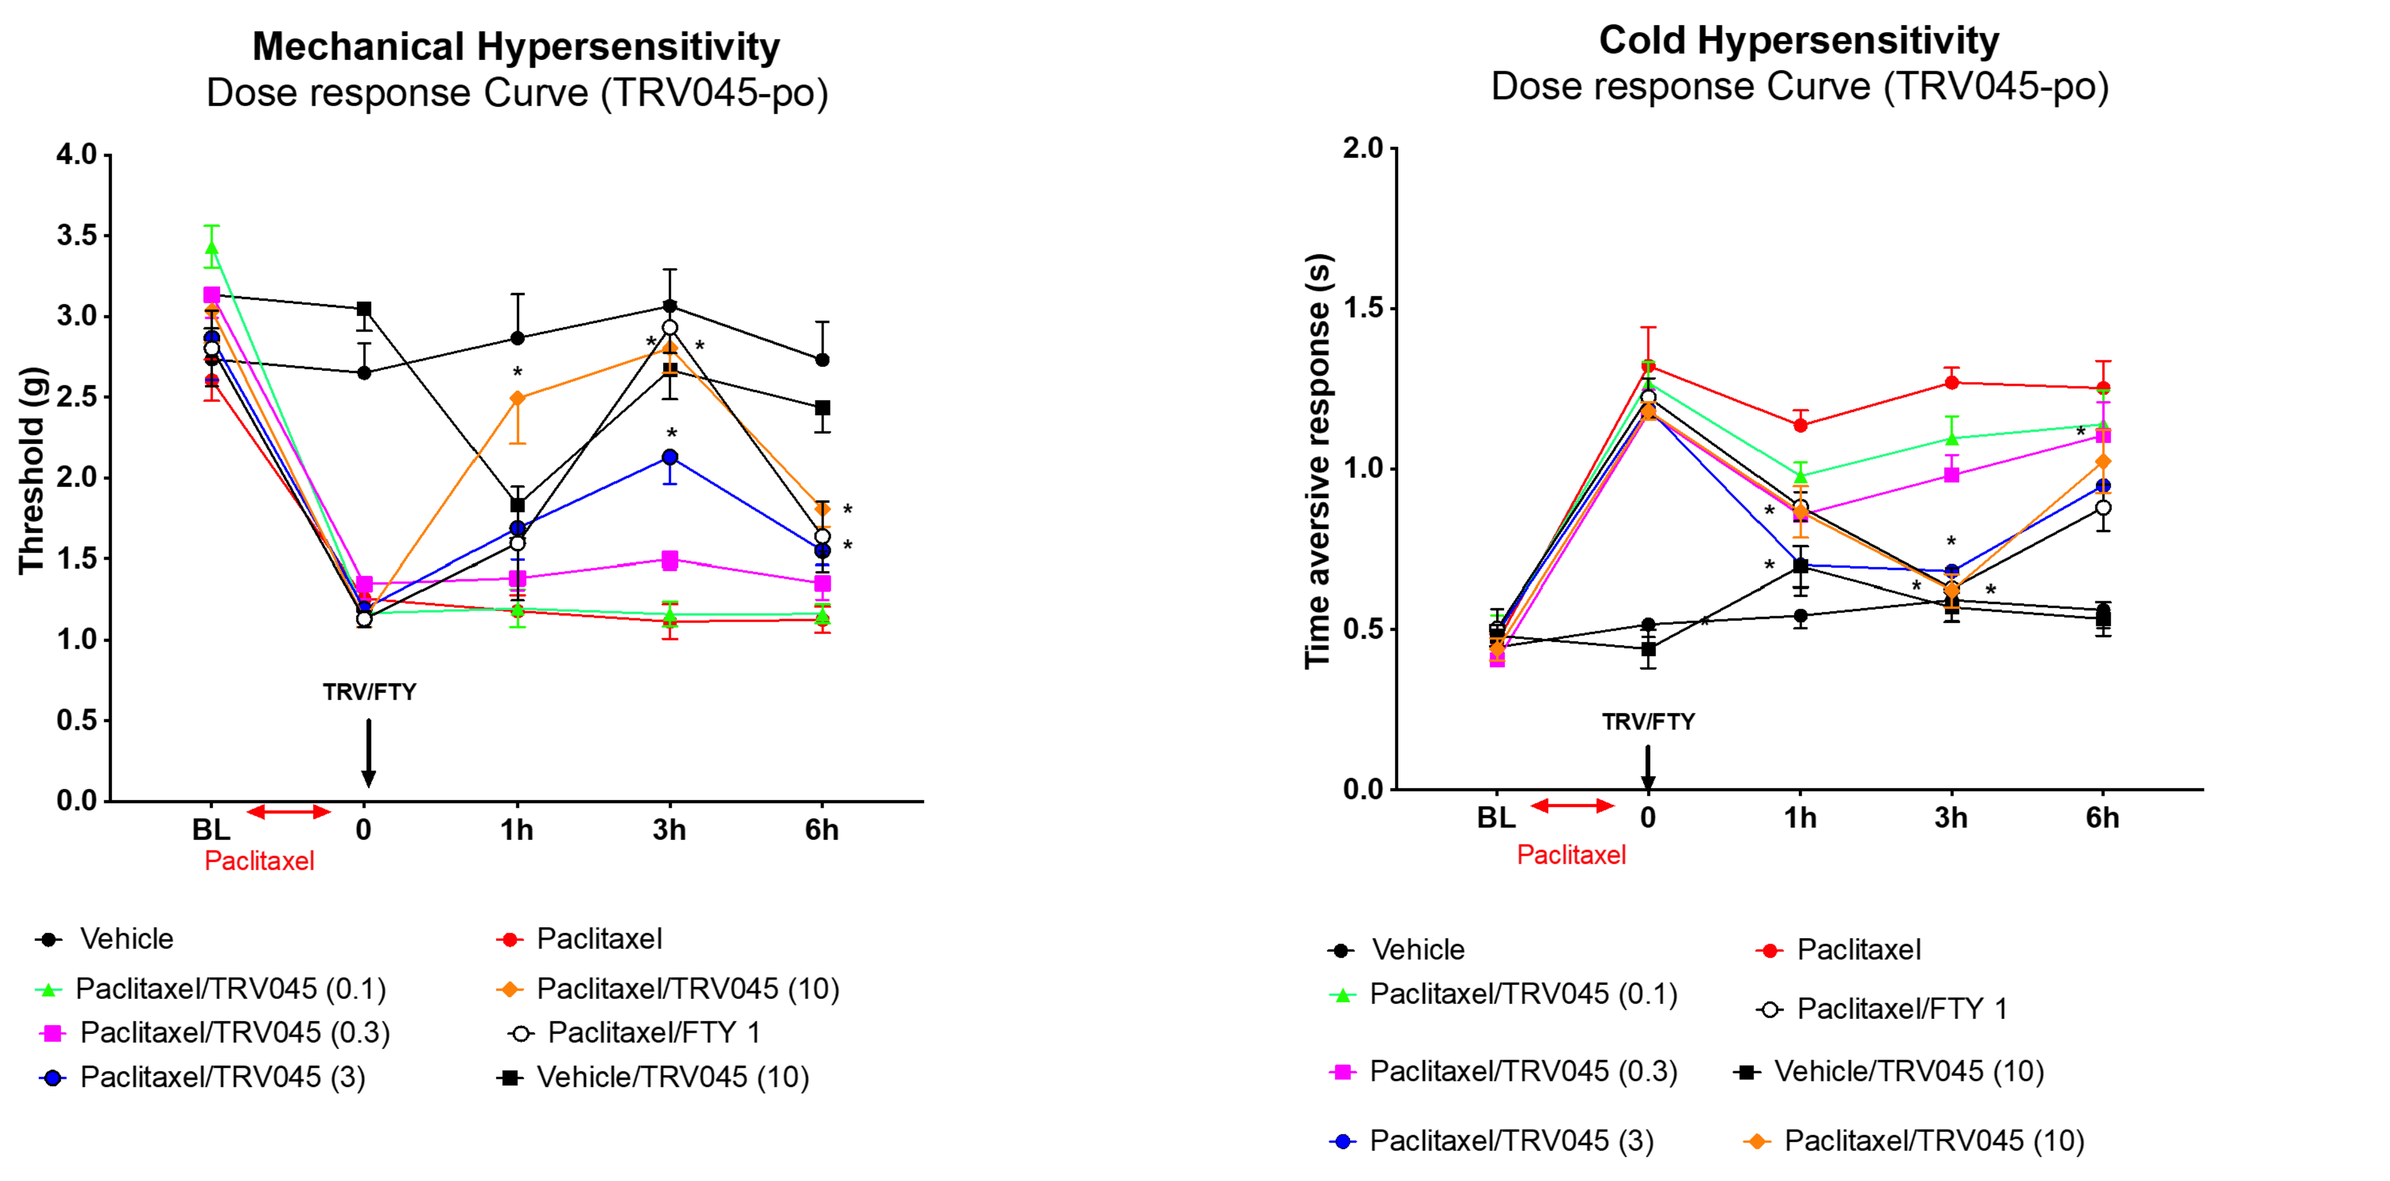
Mechanical Hypersensitivity
Dose response Curve (TRV045-po)
Threshold (g)
TRV/FTY
Paclitaxel
Cold Hypersensitivity
Dose response Curve (TRV045-po)
Time aversive response (s)
Vehicle
Paclitaxel
Paclitaxel/TRV045 (0.1)
Paclitaxel/TRV045 (0.3)
Paclitaxel/TRV045 (3)
Paclitaxel/TRV045 (10)
Paclitaxel/FTY 1
Vehicle/TRV045 (10)

NOTES: Each point represents the mean ± SEM of 8 mice per group. * Denotes significantly different from paclitaxel group.

Figure 2B. Acute Reversal of CIPN: Effect of TRV045 (females)

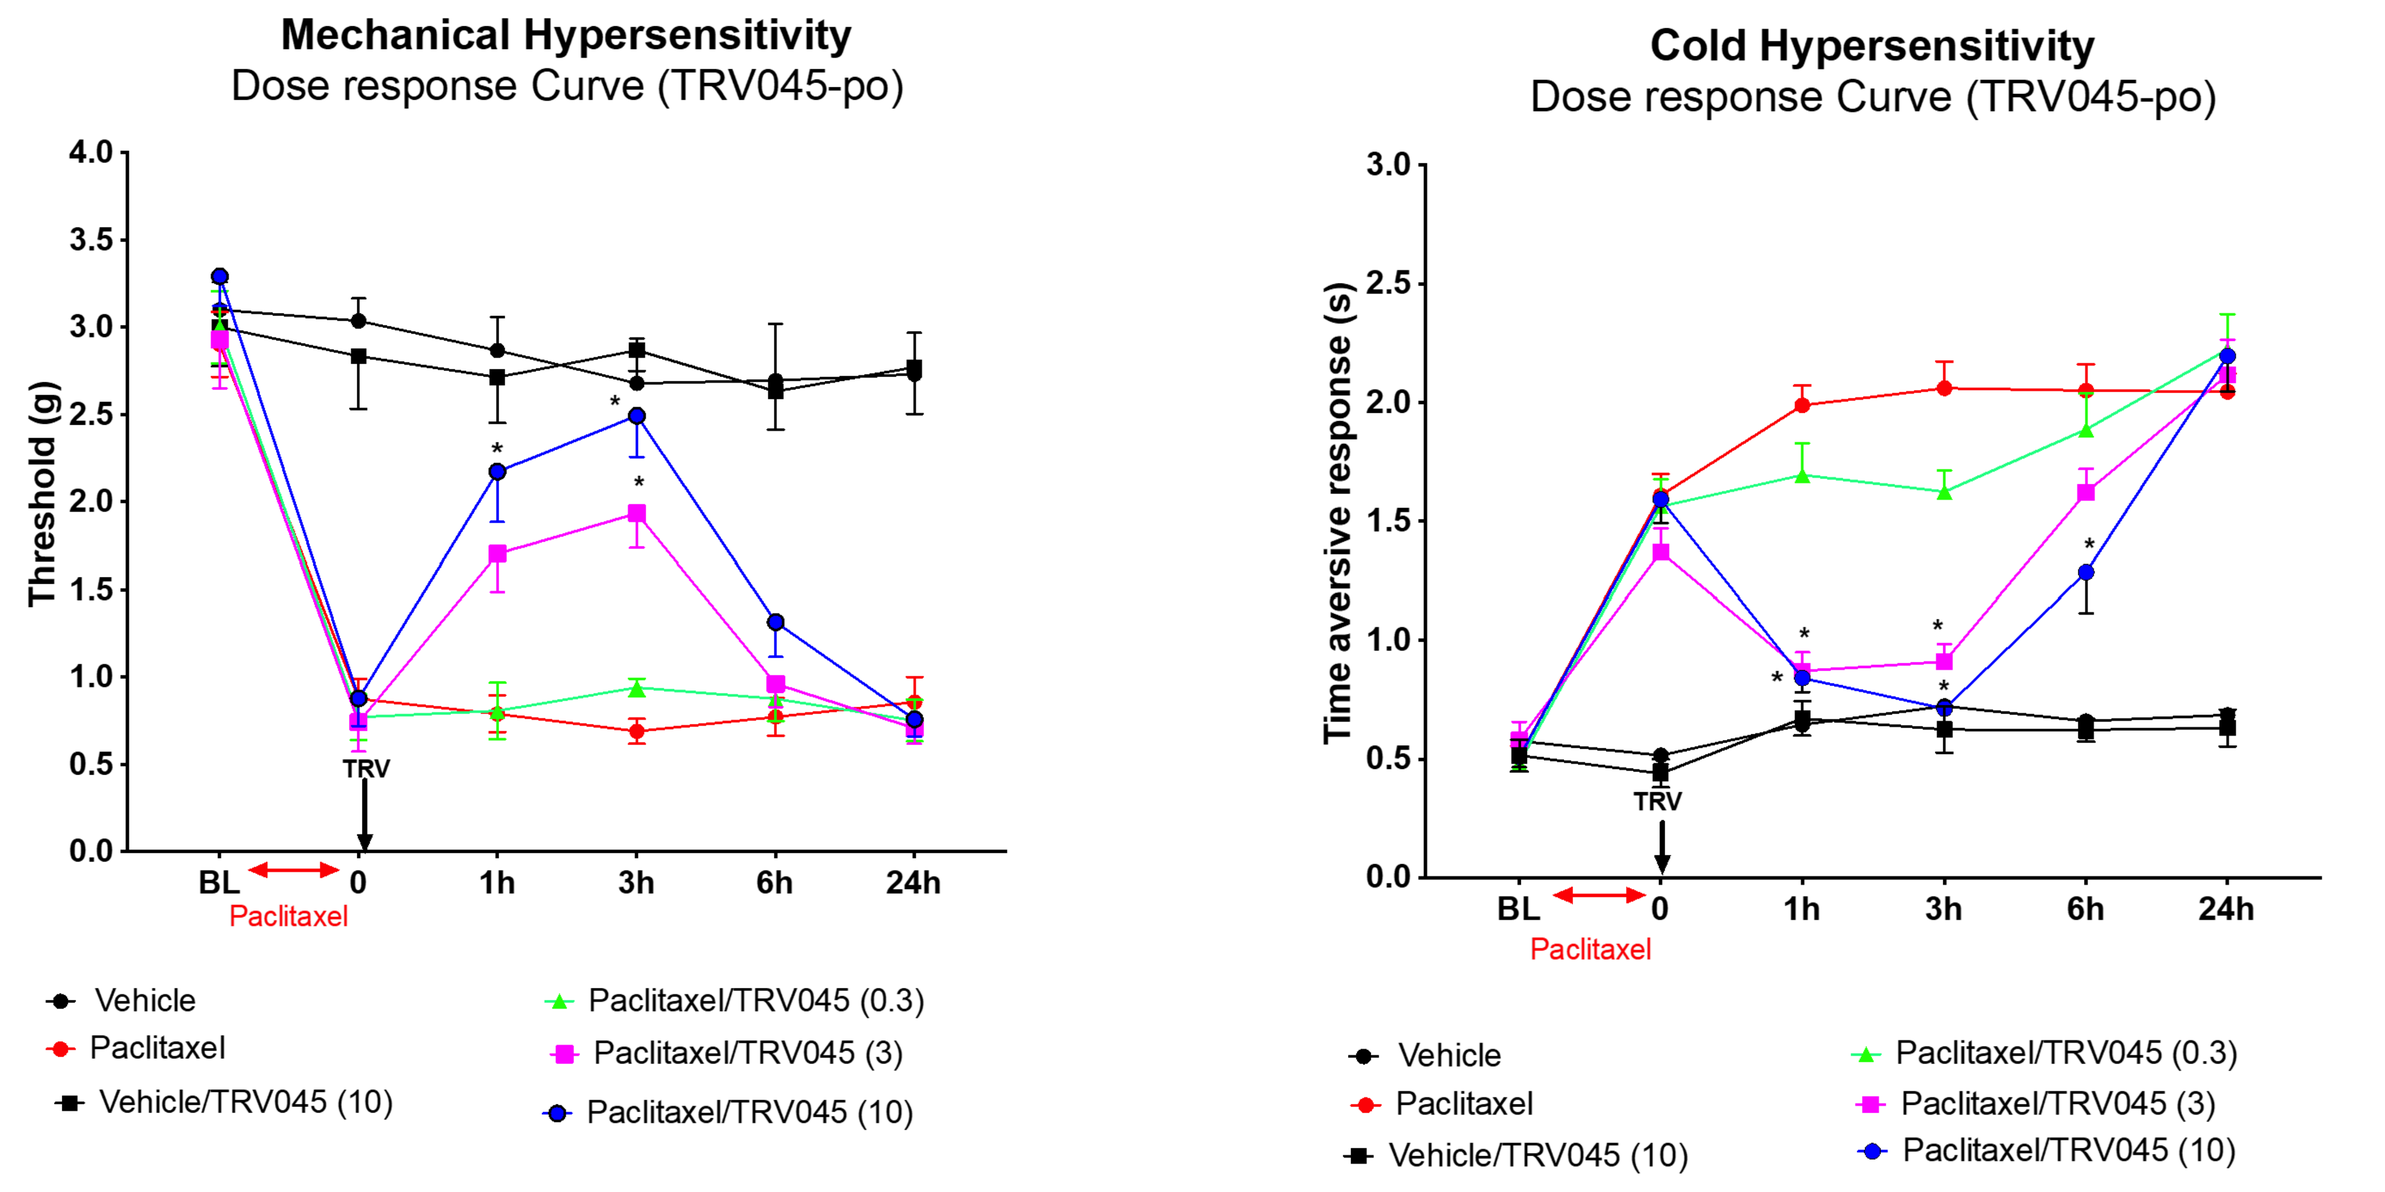
Mechanical Hypersensitivity
Dose response Curve (TRV045-po)
Threshold (g)
TRV
Paclitaxel
Cold Hypersensitivity
Dose response Curve (TRV045-po)
Time aversive response (s)
Vehicle
Paclitaxel
Vehicle/TRV045 (10)
Paclitaxel/TRV045 (0.3)
Paclitaxel/TRV045 (3)
Paclitaxel/TRV045 (10)

NOTES: Each point represents the mean ± SEM of 8 mice per group. * Denotes significantly different from paclitaxel group.

Figure 3. Sustained Reversal of CIPN: Effect of TRV045 (males)

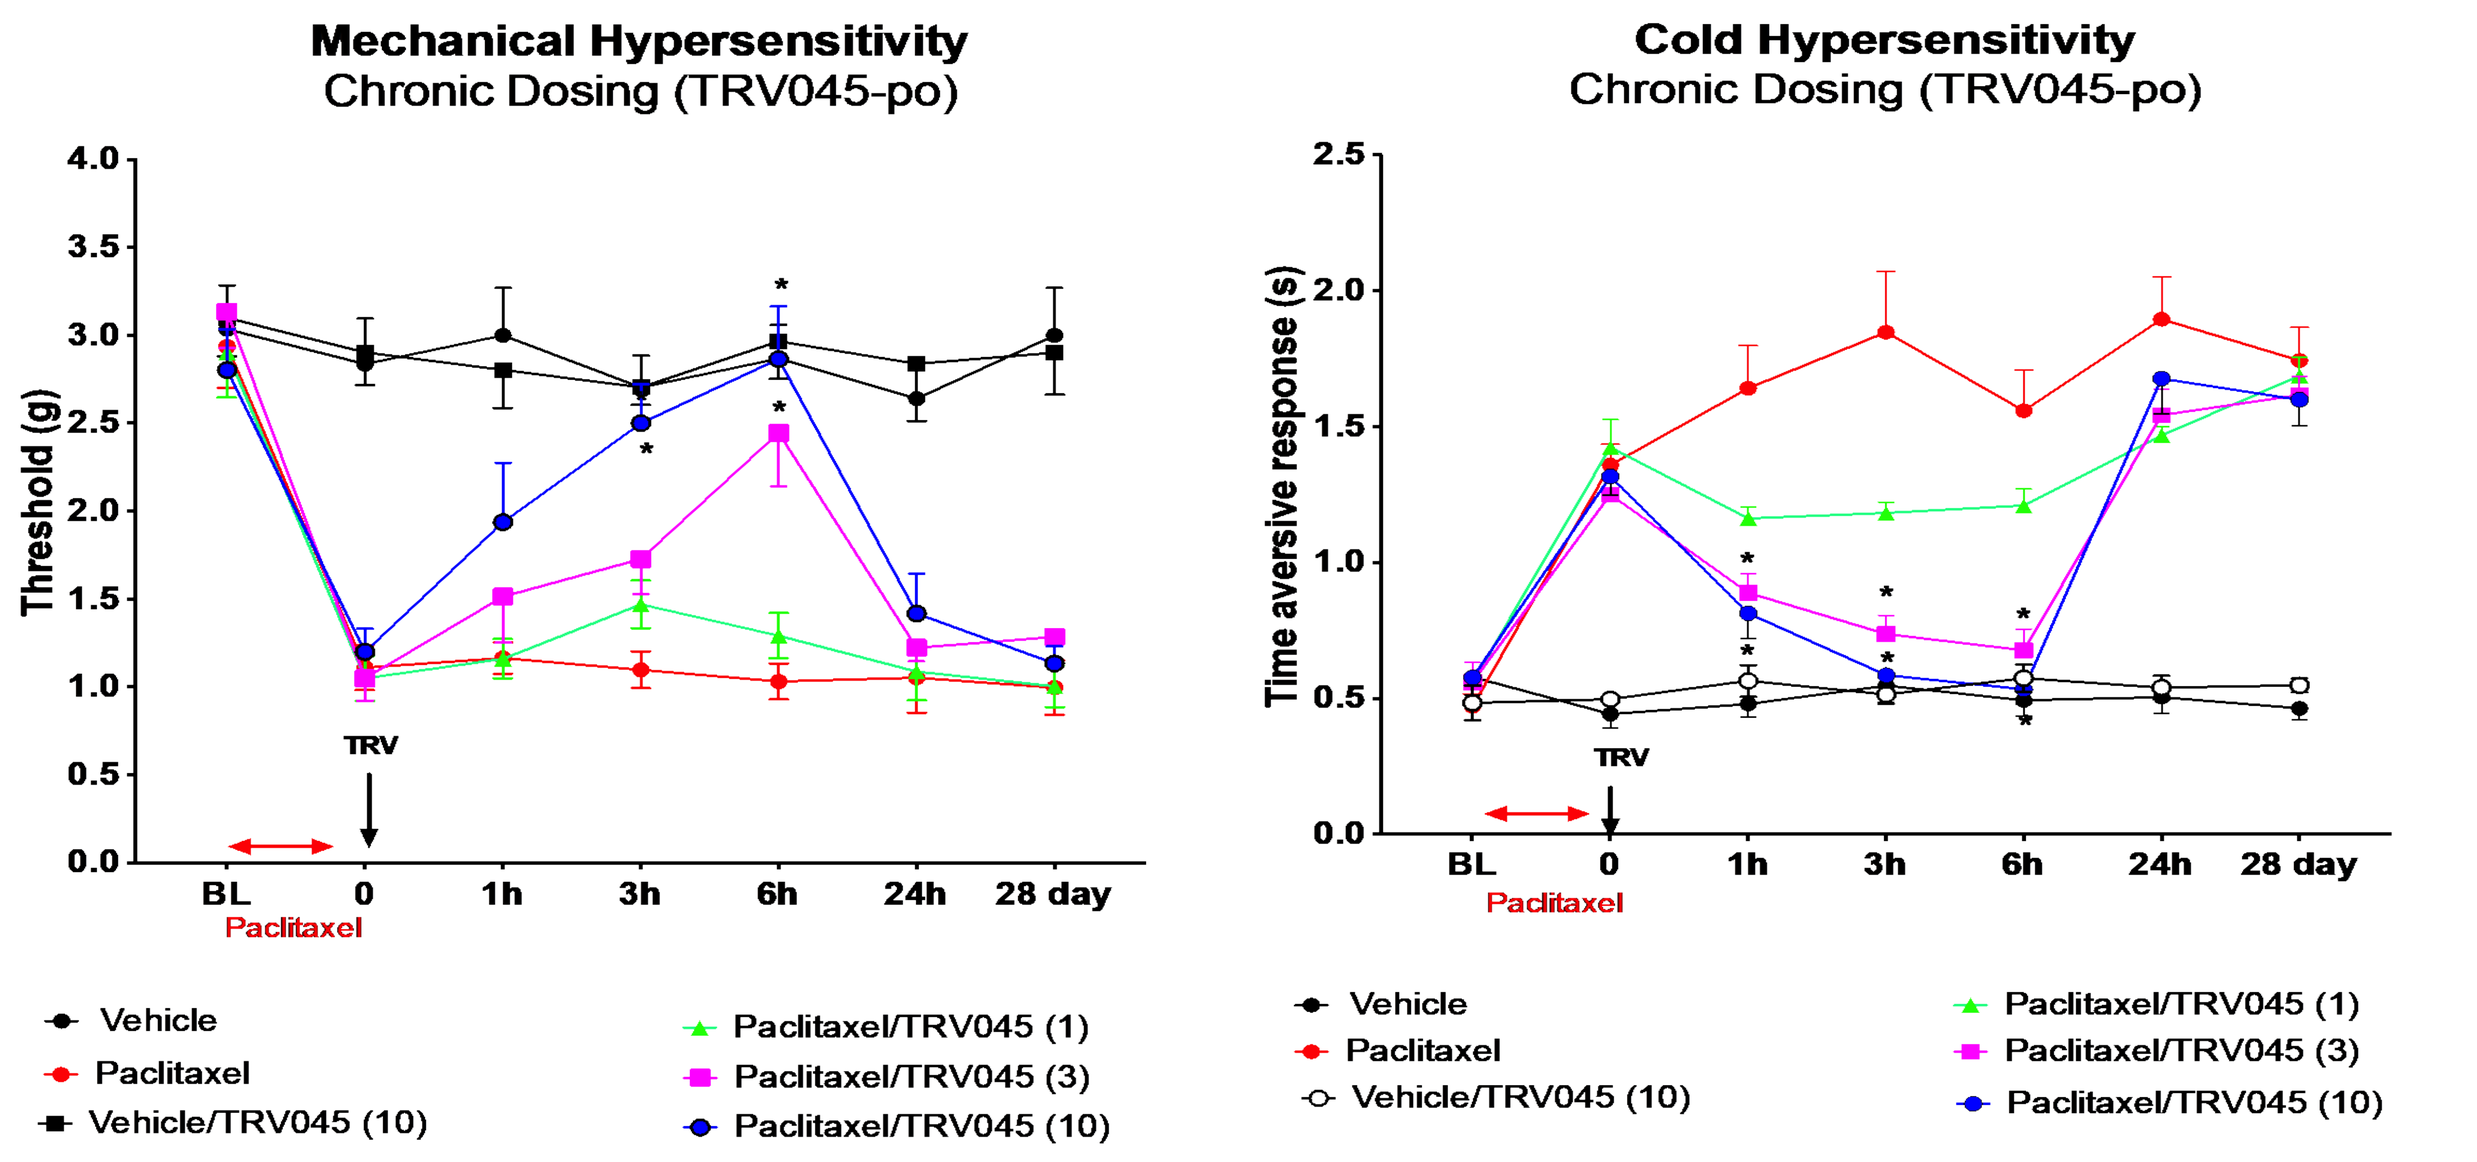
Mechanical Hypersensitivity
Chronic Dosing (TRV045-po)
Threshold (g)
TRV
Paclitaxel
Cold Hypersensitivity
Chronic Dosing (TRV045-po)
Time aversive response (s)
Vehicle
Paclitaxel
Vehicle/TRV045 (10)
Paclitaxel/TRV045 (1)
Paclitaxel/TRV045 (3)
Paclitaxel/TRV045 (10)

NOTES: Each point represents the mean ± SEM of 8 mice per group (except for the time 24 h = 4/group). * Denotes significantly different from paclitaxel group.

Figure 4. Effect of TRV045 on CIPN Prevention

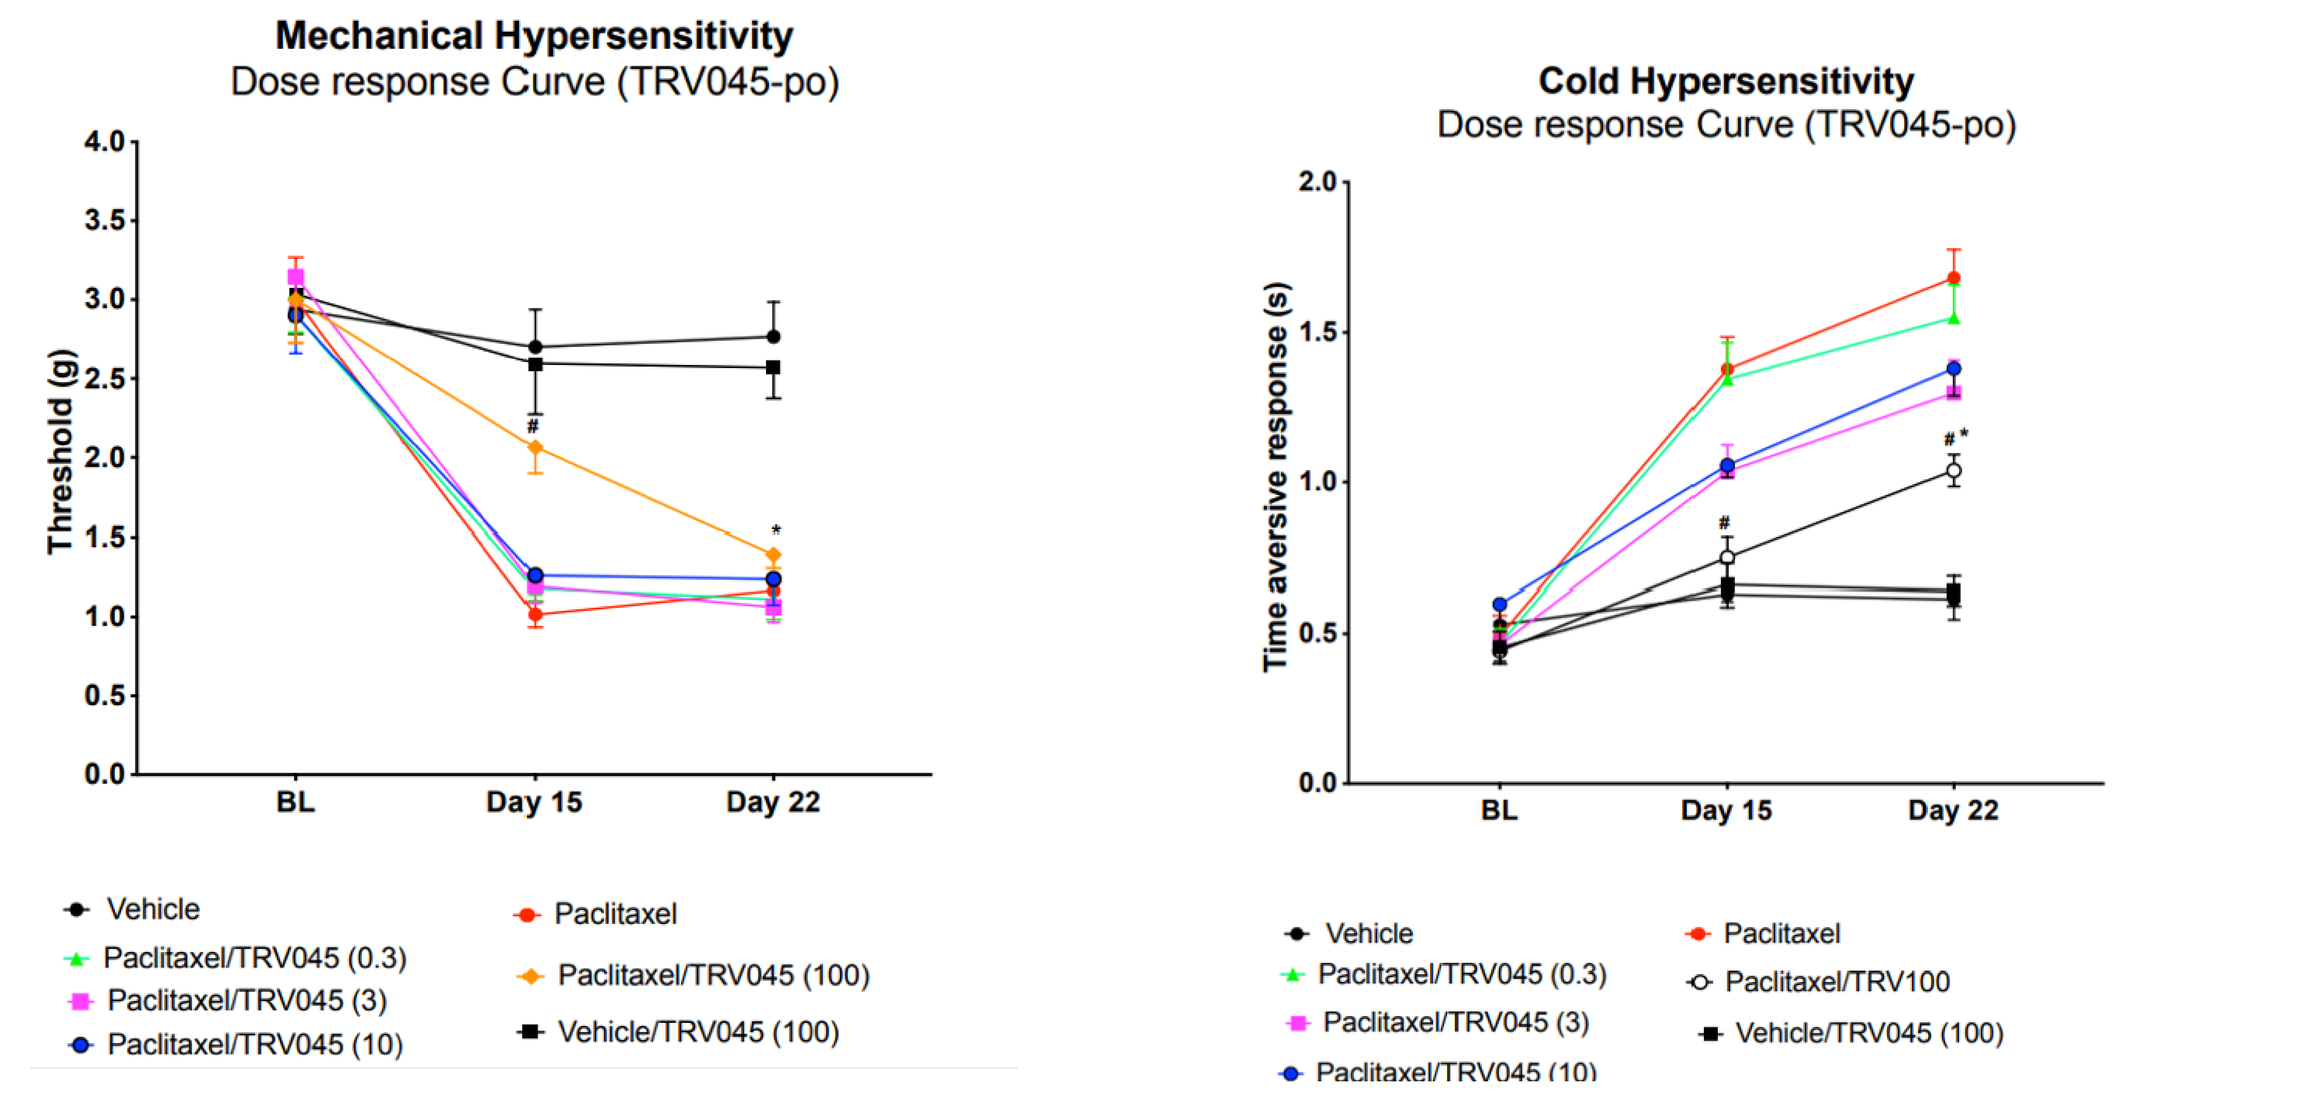
Mechanical Hypersensitivity
Dose response Curve (TRV045-po)
Threshold (g)
Cold Hypersensitivity
Dose response Curve (TRV045-po)
Time aversive response (s)
BL
Day 15
Day 22
Vehicle
Paclitaxel
Paclitaxel/TRV045 (0.3)
Paclitaxel/TRV045 (3)
Paclitaxel/TRV045 (10)
Paclitaxel/TRV045 (100)
Vehicle/TRV045 (100)
Paclitaxel/TRV100

NOTES: Each point represents the mean ± SEM of 8 mice per group. * Denotes significantly different from vehicle group; # denotes significantly different from paclitaxel group.

Figure 5. No Effect of TRV045 on Voluntary Wheel Running Behavior

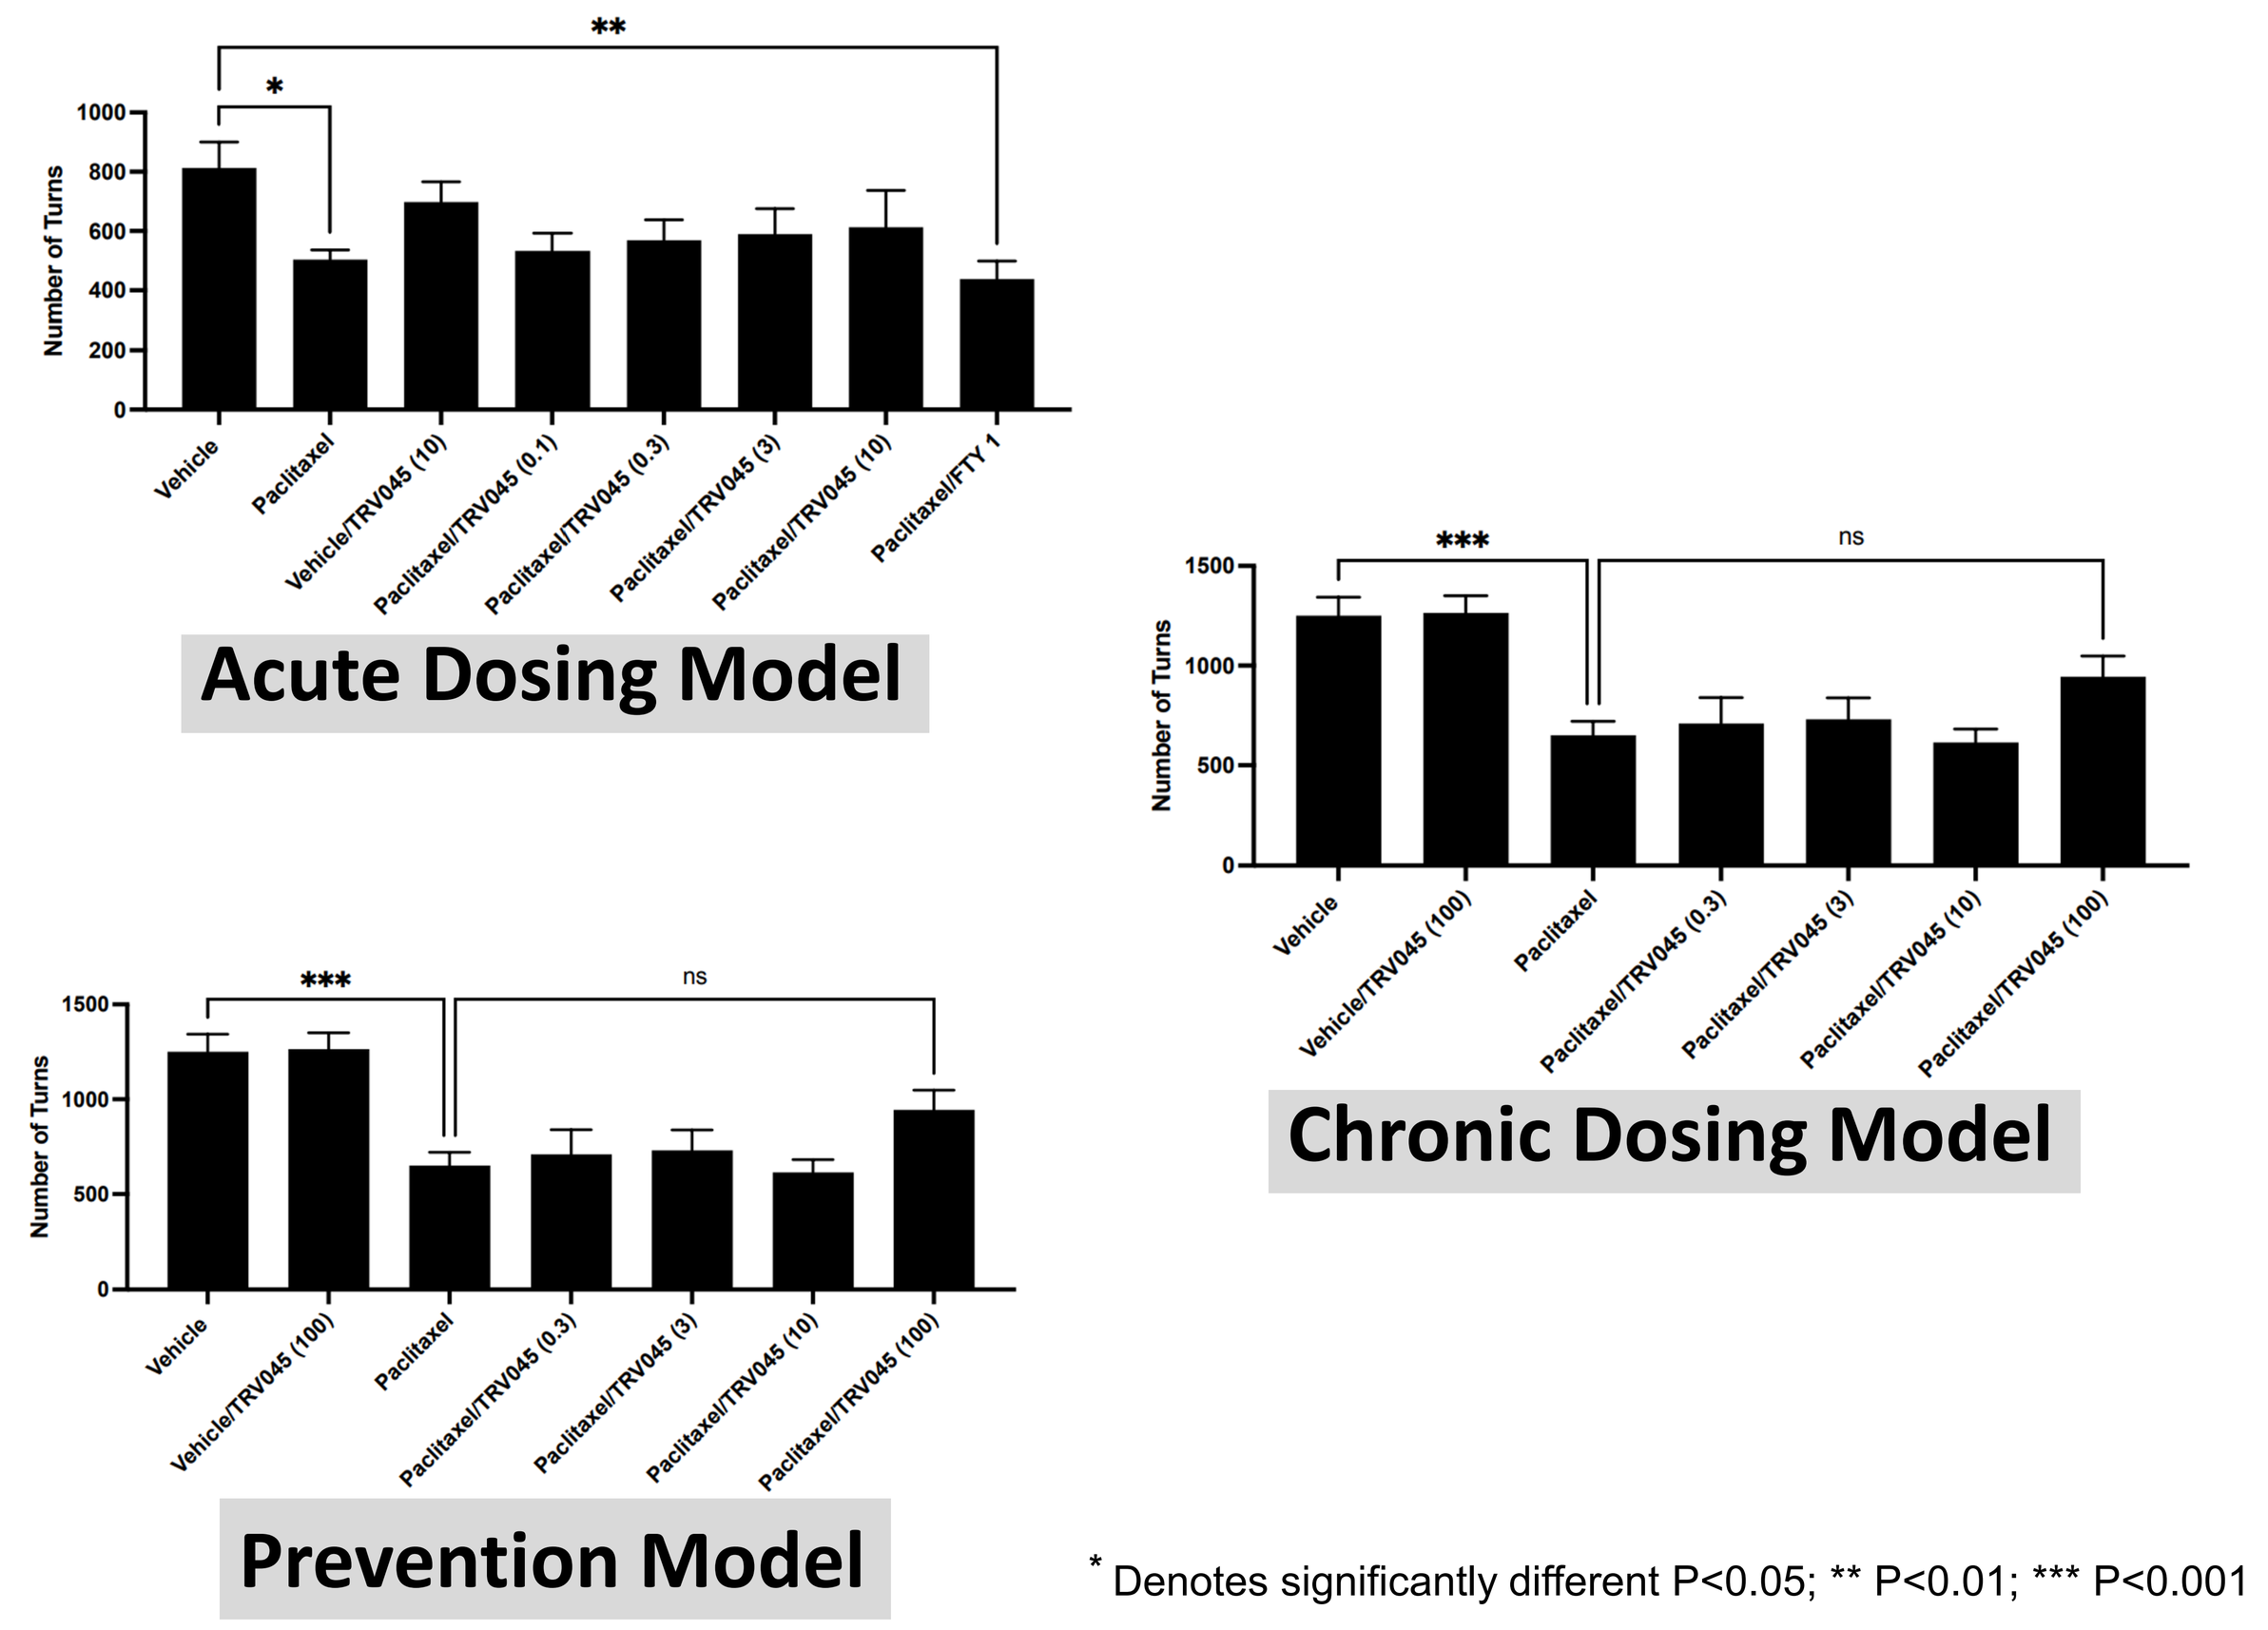
Number of Turns
Vehicle
Paclitaxel
Vehicle/TRV045 (10)
Paclitaxel/TRV045 (0.1)
Paclitaxel/TRV045 (0.3)
Paclitaxel/TRV045 (3)
Paclitaxel/TRV045 (10)
Paclitaxel/FTY 1
Acute Dosing Model
Vehicle/TRV045 (100)
Paclitaxel/TRV045 (100)
ns
Chronic Dosing Model
Prevention Model

* Denotes significantly different P<0.05; ** P<0.01; *** P<0.001

CONCLUSIONS

- Oral administration of TRV045 reduced mechanical and cold stimulus-evoked nociception in a dose-related manner in a mouse model of CIPN
- These effects were demonstrated in single-dose acute reversal and were sustained following repeated administration of TRV045 for 14 days; effects were evident at doses of 3mg/kg and 10mg/kg
- In a prevention mode paradigm examining the potential effect of TRV045 to prevent the development of CIPN, TRV045 administered at 100mg/kg reduced both mechanical and cold hypersensitivity 24 hours following the last dose; the reduction in cold hypersensitivity was present at 168 hours after the last dose of TRV045

REFERENCES

1. Caillaud, M., et al, 2021 *Brain Behav Immun. 93: 172–185*
2. Coste et al, 2008, *J Cell Mol Med.* 12(3):995-1004
3. Doolen et al, 2018, *Pain* 159(2):224-238
4. Sim-Selley et al, 2018, *J Pharmacol Exp Ther.* 366(3):509-518
5. Yamazaki et al, 2020, *J Neurotrauma* 37(15):1720-1728
6. Squillace et al, 2020, *Trends Pharmacol Sci.* 41(11):851-867

Disclosure: TRV045 is an investigational new drug that has not been approved by the FDA.

Presented at the 62nd annual meeting of The American College of Neuropsychopharmacology (ACNP), December 3-6, 2023