T158

Oliceridine demonstrates a reduced effect on neurocognitive function in humans, compared to morphine: A Phase 1, randomized, placebo-controlled, dose-ranging, partial block, cross-over study

Mark A. Demitrack, Michael S. Kramer, Cathy Michalsky, Hemme Hijma, Laurence Moss, Geert Jan Groeneveld; Trevena, Inc., Chesterbrook, PA, Centre for Human Drug Research, Leiden, Netherlands

BACKGROUND

Oliceridine is an agonist at the mu-opioid receptor, with preferential post-receptor engagement of G-protein signaling, while showing reduced beta-arrestin recruitment and receptor internalization (DeWire, 2013).

Opioid medications are an essential component of pain management after surgery, though a variety of opioid-induced adverse events (AEs) complicate their use. Notable among these is the development or exacerbation of cognitive dysfunction, which may range from sedation to confusion or progress to delirium. Cognitive dysfunction can therefore have potential implications for post-operative recovery and health outcomes, and in some instances may result in deficits that persist beyond the immediate post-operative period. The mechanism of these cognitive complications is unclear, though it has been hypothesized that opioids, such as morphine, can bind to the toll-like receptor 4 (TLR4), and the subsequent neuroinflammatory response may contribute to these postoperative cognitive sequelae (Muscat, 2021). Rats treated with oliceridine demonstrate reduced levels of spinal cord TLR4 after experimental fracture compared to morphine-treated animals (Liang, 2018). The present study was designed to characterize the neurocognitive impact of IV oliceridine versus IV morphine using a validated cognitive test battery. We hypothesized that IV oliceridine would demonstrate a reduced effect on cognitive measures compared to IV morphine.

OBJECTIVE

Evaluate the comparative neurocognitive profile of oliceridine and morphine across a therapeutically relevant dose range in healthy volunteers.

METHODS

Twenty-three healthy subjects (13 males, 10 females; median age 26 years), provided informed consent and were randomized to 3 of the 5 possible treatments as single IV doses in a partial block cross-over design: placebo, oliceridine 1 mg or 3 mg, or morphine 5 mg or 10 mg. The dose ranges were selected based on prior data confirming a relative potency of oliceridine to morphine of approximately 1 to 5, and the maximum approved single dose of oliceridine (3 mg; Olinvyk® product label).

Neurocognitive function using the NeuroCart® test battery was assessed at: baseline, 30 min, 1h, 2h, 3h, 4h, 5h and 6h. The primary outcome of the study was saccadic eye movement peak velocity, a sensitive measure of sedation. Secondary outcome measures included saccadic eye movement reaction time and % inaccuracy, smooth pursuit eye movement, an adaptive tracking test of eye-hand coordination, postural stability measured by body sway, and the symbol-digit substitution test. Right and left pupillometry, and analgesia measured with the cold pain test assessed adequacy of target engagement. Outcomes were examined with a mixed-model ANOVA, with treatment, period, time, and treatment by time as fixed factors, subject, subject by treatment and subject by time as random factors and average baseline value as a covariate. For a significant main effect of treatment, between-group pairwise comparisons of the treatment conditions were performed.

RESULTS

Consistent with the known relative potency of IV oliceridine and IV morphine, across the dose ranges studied, both drugs demonstrated expected effects on opioid-induced pupillary constriction (data not shown), and analgesia in response to cold pain testing (Figure 1).

There was a statistically significant effect of treatment on the primary outcome measure of saccadic eye movement peak velocity (main effect of treatment, P<0.0001), driven by a favorable, reduced impact of oliceridine versus IV morphine (LS mean treatment difference [95% CI]: -11.40 degrees/s [-21.19, -1.61], P=0.0236) (Figure 2). Similar outcomes favoring oliceridine were observed on the secondary outcome measures of saccadic eye movement reaction time (main effect of treatment, P=0.0201; LS mean treatment difference [95% CI]: 0.0088 sec [0.0010, 0.0166], P=0.0273), reduced body sway (main effect of treatment, P=0.0314; LS mean treatment difference [95% CI]: 16.1% (mm) [-2.7%, 38.5%], P=0.0951) (Figure 3), and improved performance accuracy on the adaptive tracking test, a measure of eye-hand coordination and sustained attention (main effect of treatment, P=0.0011; LS mean treatment difference [95% CI]: -1.519% [-3.505, 0.467], P=0.1303) (Table 1).

Additional neurocognitive outcome measures, visual tracking and the symbol-digit substitution test, did not show statistical differences between oliceridine and IV morphine (Table 1). No serious adverse events were observed in the study. Common AEs included expected opioid-related adverse events of nausea, vomiting and somnolence. Events were assessed as mild in all cases with the exception of one AE of moderate nausea in a morphine-treated subject.

Figure 1. IV Olicerdine and IV Morphine Cold Pain Tolerance (sec)

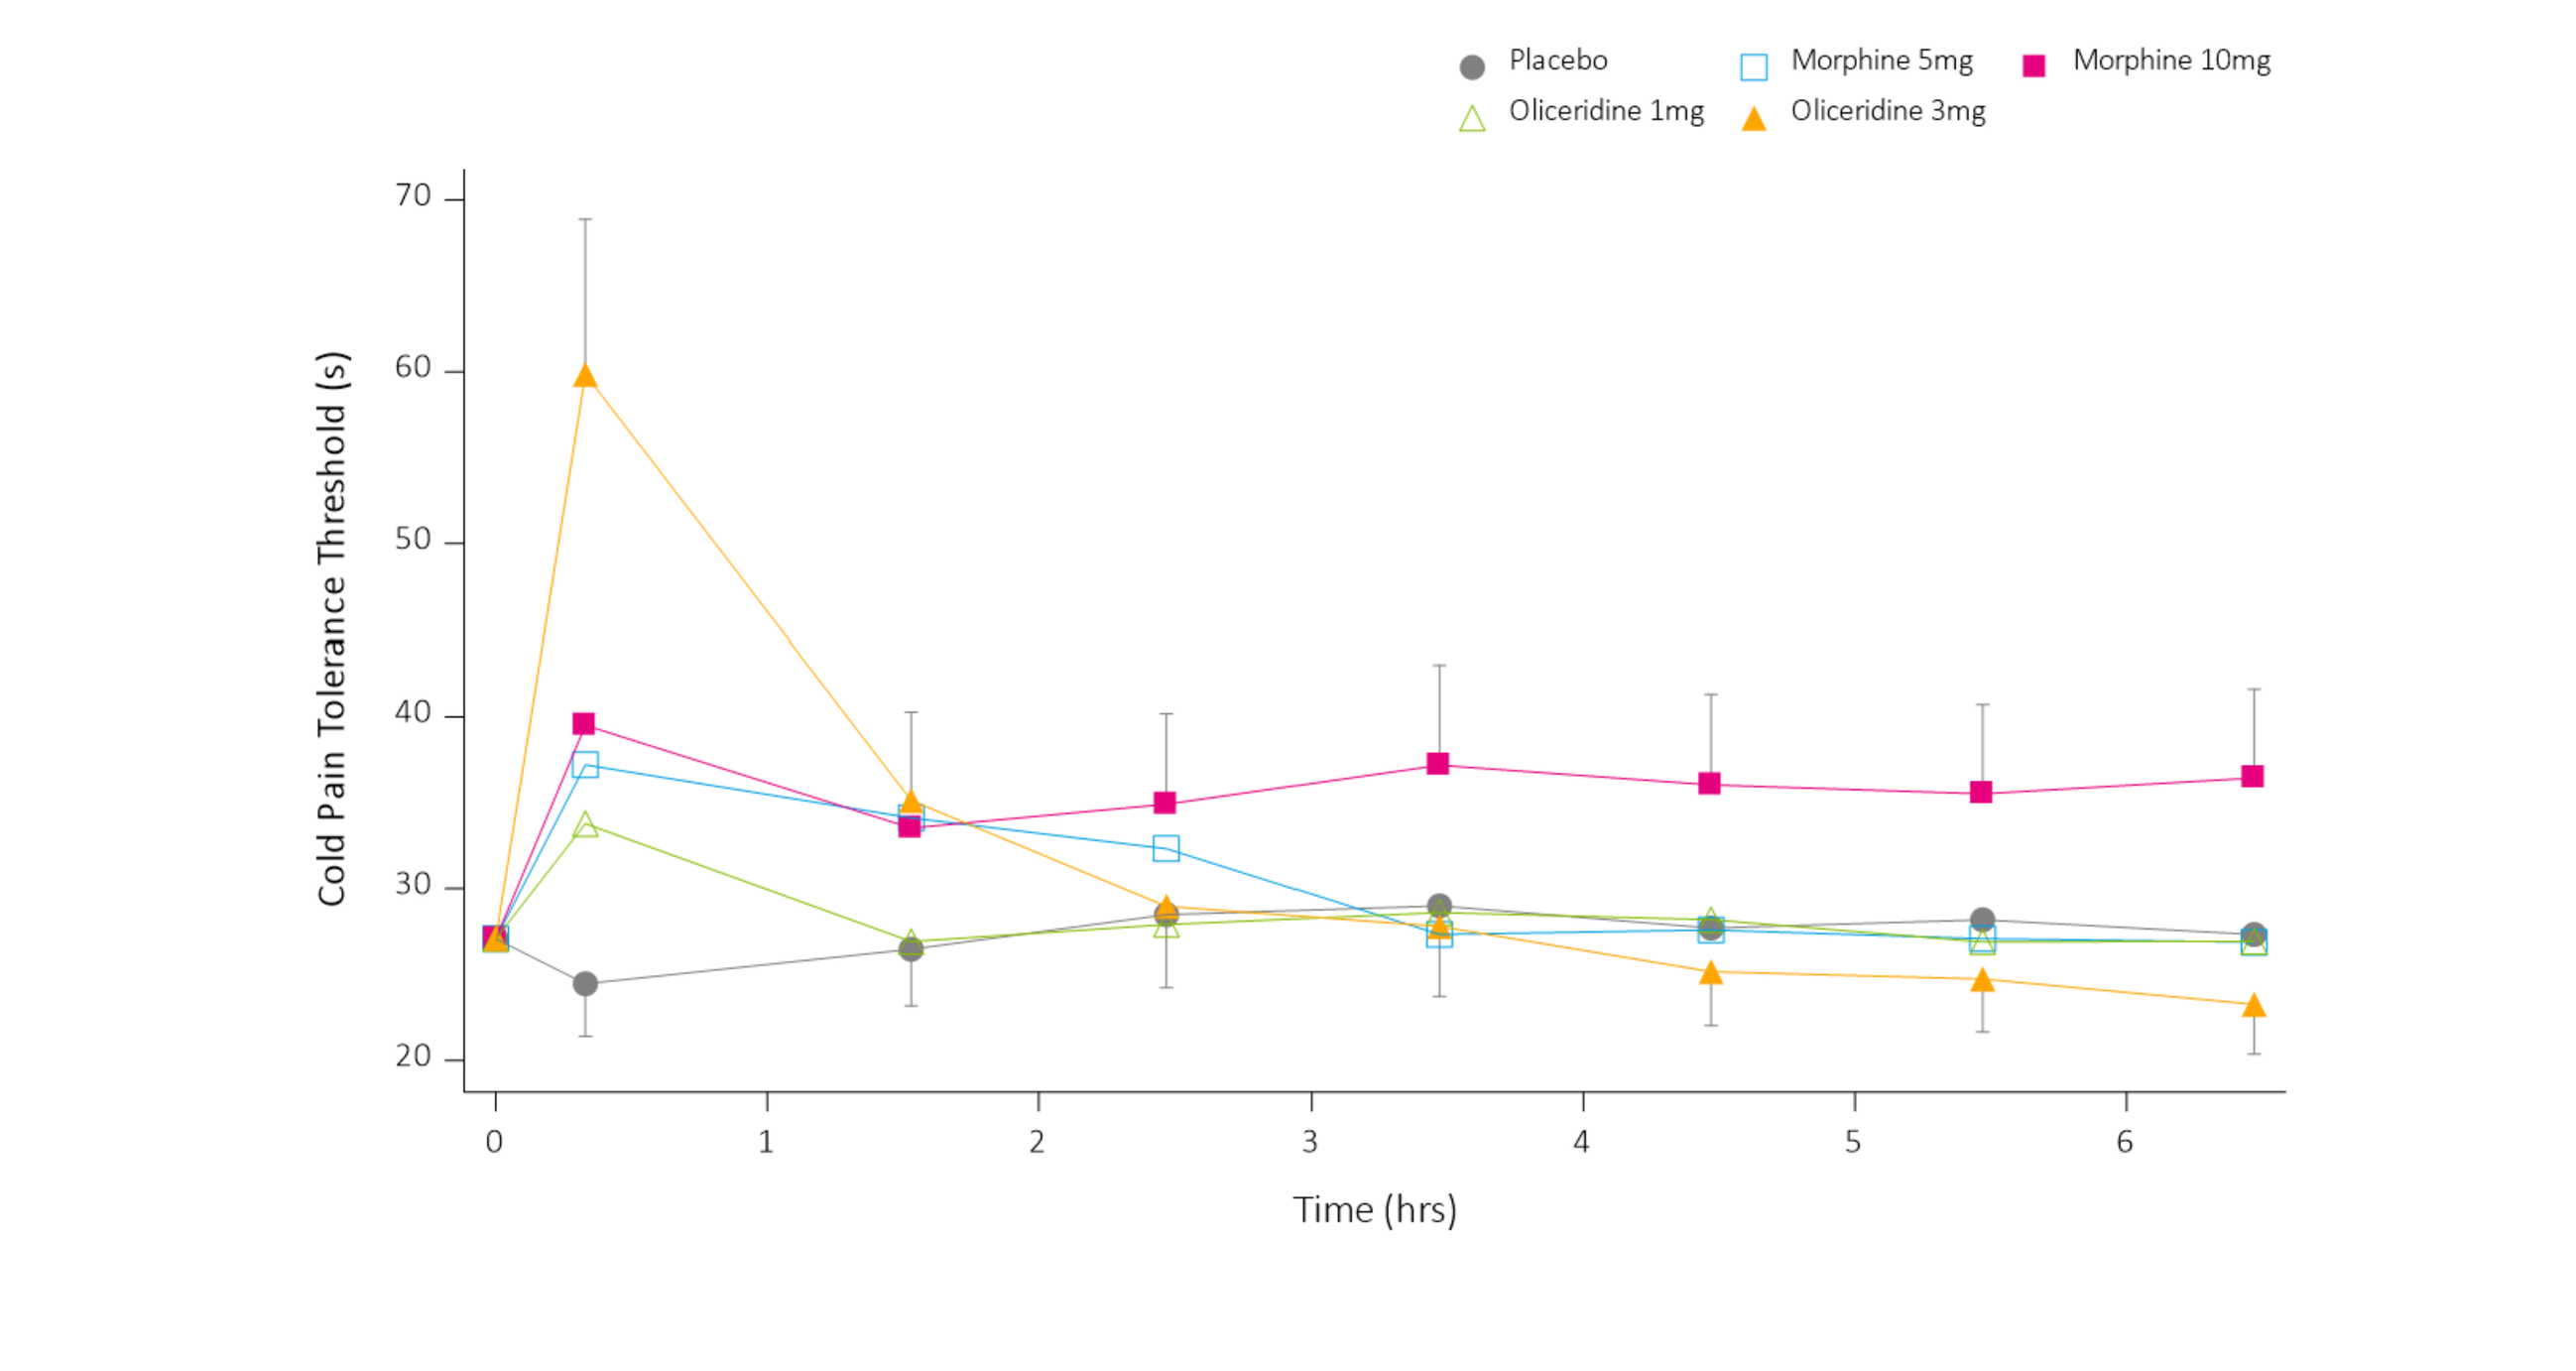
Placebo
Morphine 5mg
Morphine 10mg
Oliceridine 1mg
Oliceridine 3mg
Cold Pain Tolerance Threshold (s)
70
60
50
40
30
20
0
1
2
3
4
5
6
Time (hrs)

Statistical analysis:(main effect of treatment, P=0.0004; LS change from baseline [95% CI]: 12.1% [3.2%, 21.9%], P=0.0236), favoring morphine

Figure 2. Primary Outcome: IV Oliceridine and IV Morphine Effects on Saccadic Peak Velocity (degrees/sec)

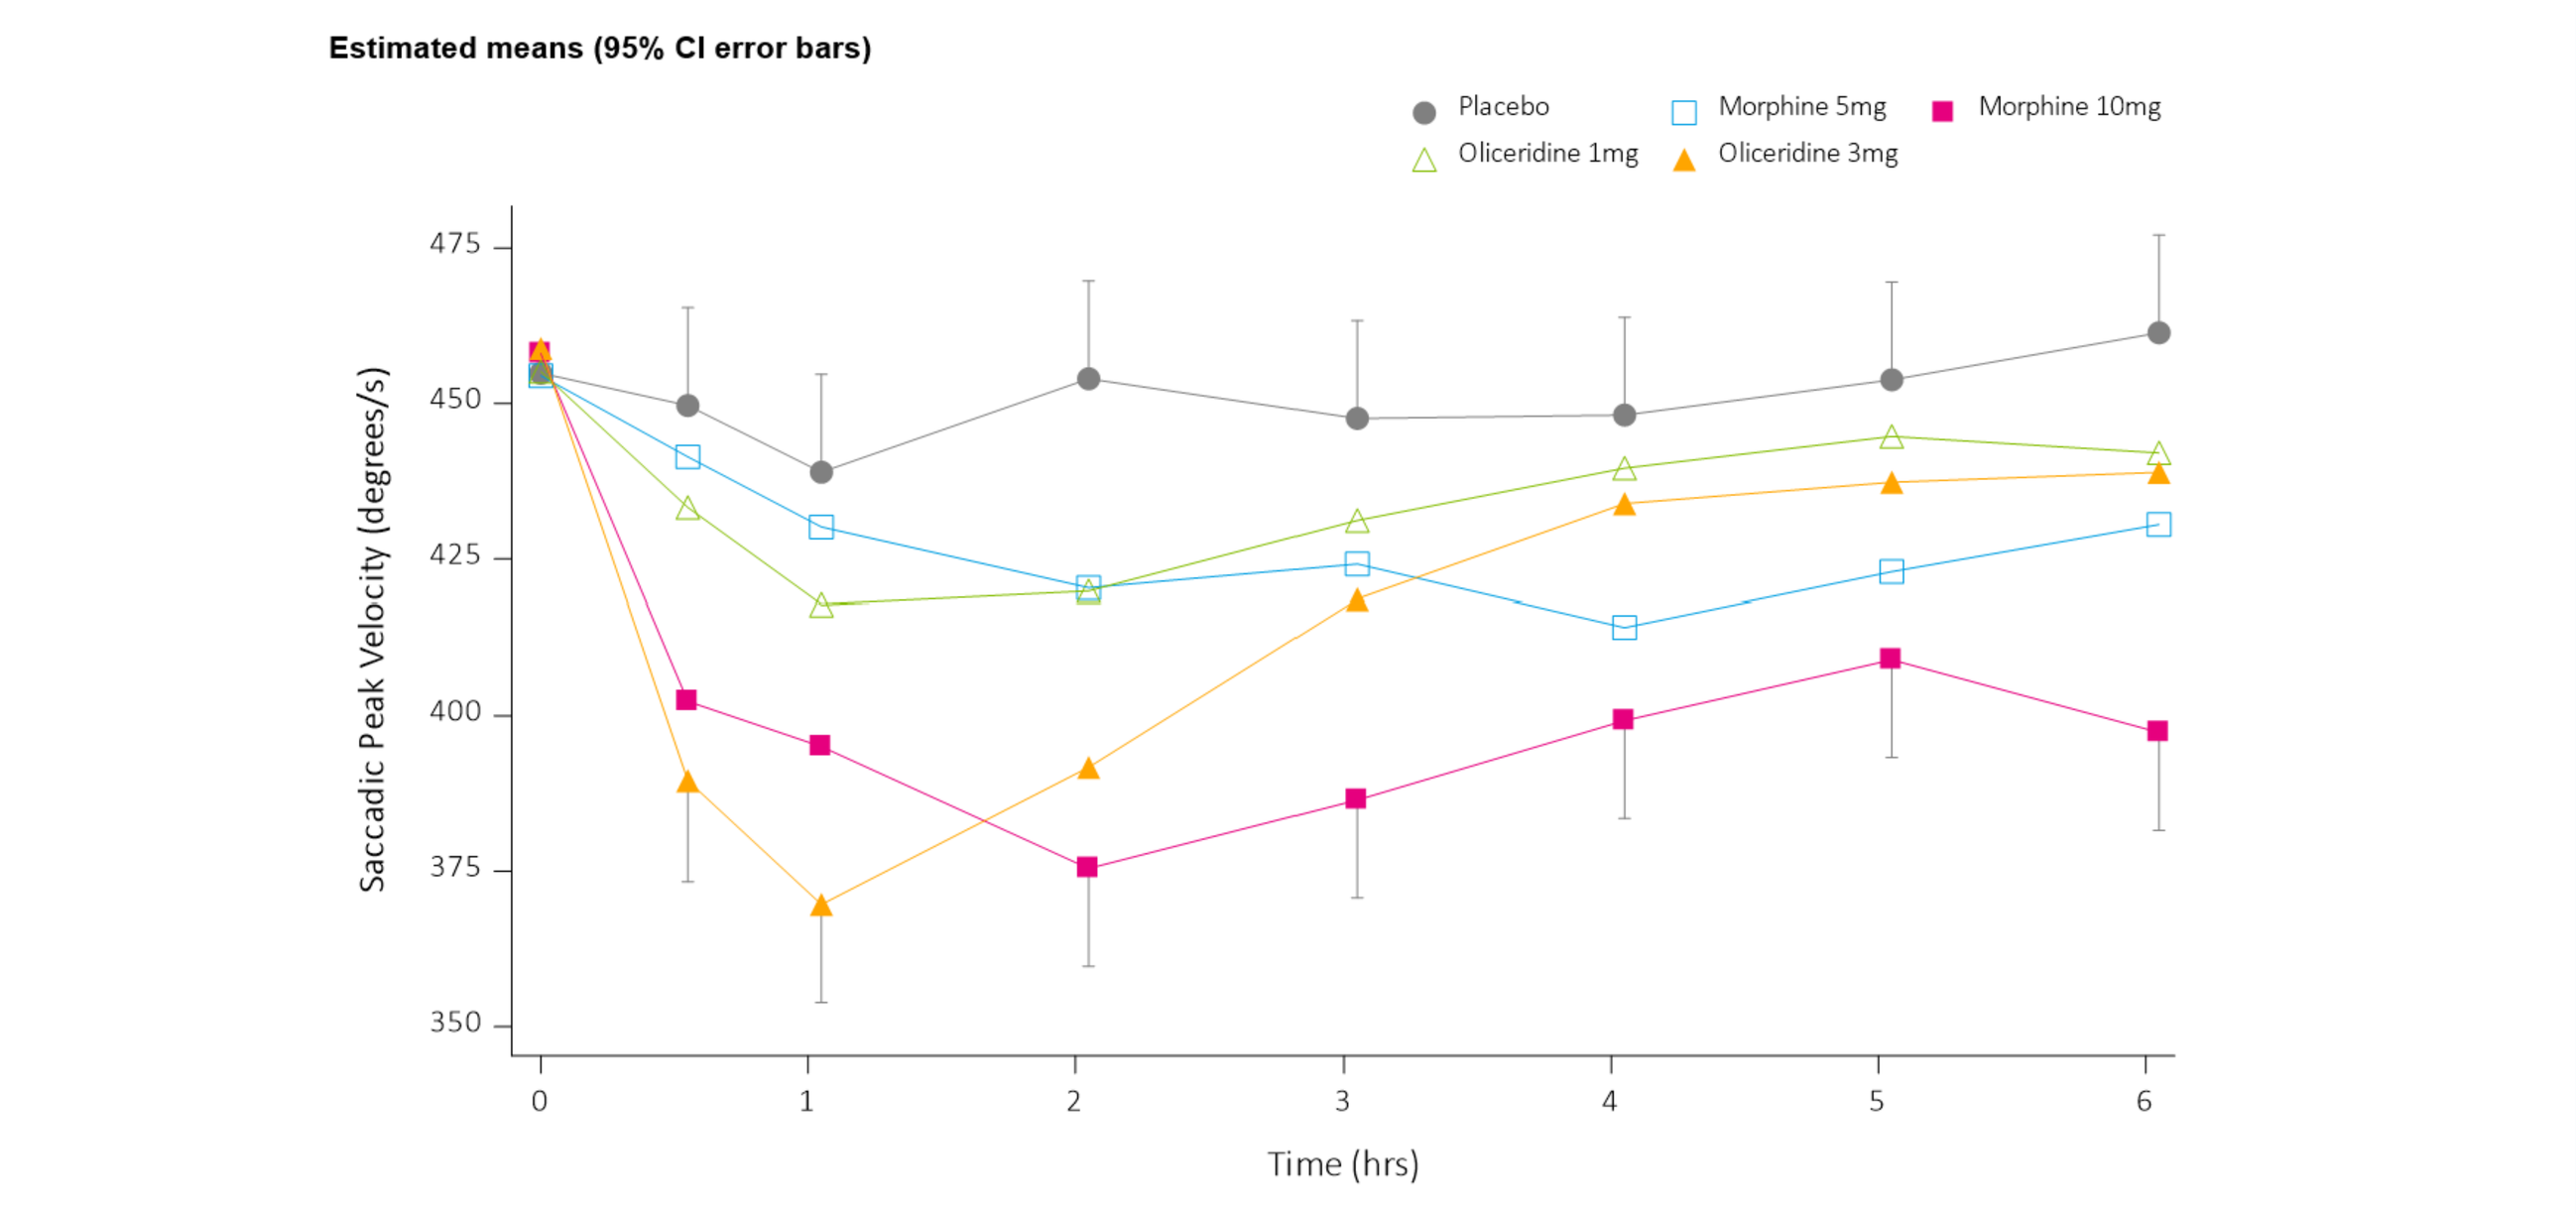
Estimated means (95% CI error bars)
Placebo
Morphine 5mg
Morphine 10mg
Oliceridine 1mg
Oliceridine 3mg
Saccadic Peak Velocity (degrees/s)
475
450
425
400
375
350
0
1
2
3
4
5
6
Time (hrs)

Statistical analysis: (main effect of treatment, P<0.0001; LS mean treatment difference [95% CI]: -11.40 degrees/s [-21.19, -1.61], P=0.0236), favoring oliceridine

Figure 3. Secondary Outcome: Body Sway (mm)

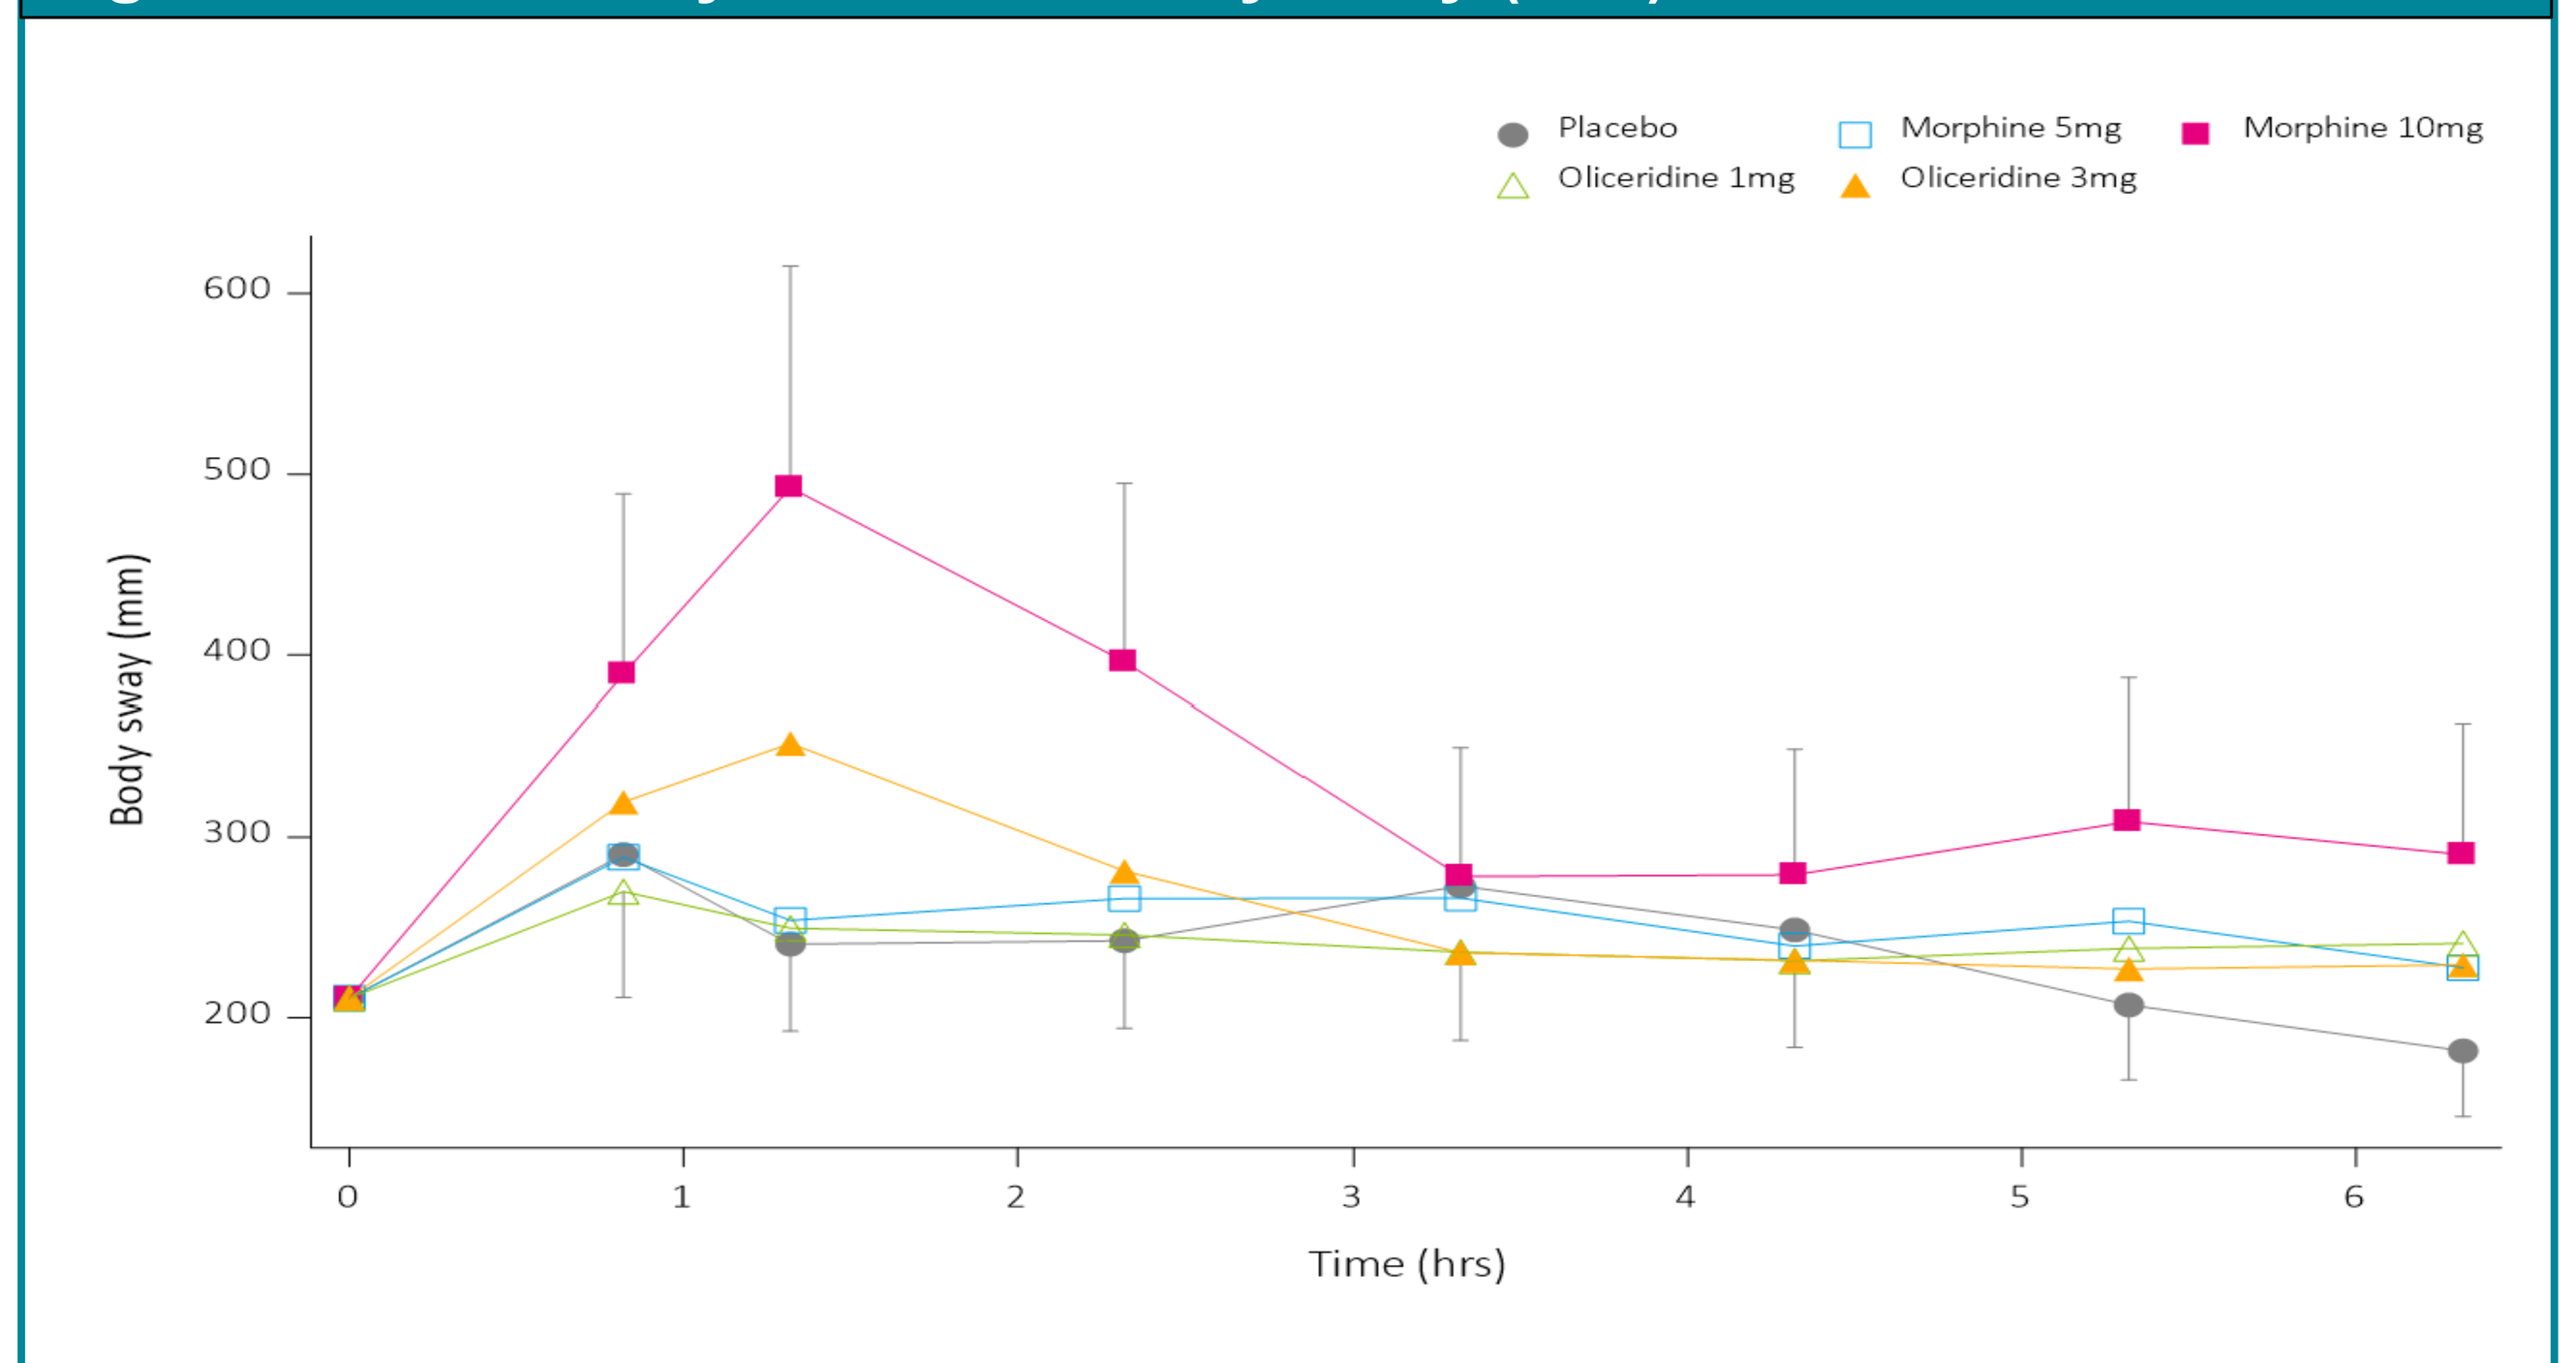
Placebo
Morphine 5mg
Morphine 10mg
Oliceridine 1mg
Oliceridine 3mg
Body sway (mm)
600
500
400
300
200
0
1
2
3
4
5
6
Time (hrs)

Statistical analysis: (main effect/treatment, P=0.0314; LS mean treatment difference [95% CI]: 16.1% (mm) [-2.7%, 38.5%], P=0.0951)

Table 1. Secondary Outcomes: Summary of Analysis Results

Parameter	Treatment P-Value	Morphine 5mg v Pbo	Morphine 10mg v Pbo	Oliceridine 1mg v Pbo	Oliceridine 3mg v Pbo	Oliceridine v Morphine
Saccadic Reaction Time (sec)	0.0201	0.0050 (-0.0059, 0.0159) p=0.3576	0.0164 (0.0056, 0.0271) p=0.0038	-0.0009 (-0.0118, 0.0100) p=0.8660	0.0047 (-0.0061, 0.0155) p=0.3838	0.0088 (0.0010, 0.0166) p=0.0273
Saccadic Inaccuracy (%)	0.0356	0.14 (-0.61, 0.88) p=0.7150	1.05 (0.32, 1.78) p=0.0064	0.43 (-0.32, 1.17) p=0.2505	0.80 (0.06, 1.54) p=0.0341	-0.02 (-0.55, 0.51) p=0.9399
Smooth Pursuit (%)	0.9287	0.44 (-3.78, 4.66) p=0.8329	1.29 (-2.84, 5.42) p=0.5295	1.59 (-2.59, 5.77) p=0.4449	0.34 (-3.79, 4.48) p=0.8667	-0.10 (-3.10, 2.90) p=0.9463
Body Sway (mm)	0.0314	7.5% (-15.7%, 37.0%) p=0.5513	42.9% (12.7%, 81.3%) p=0.0040	2.7% (-19.6%, 31.1%) p=0.8292	11.0% (-13.3%, 42.1%) p=0.3989	16.1% (-2.7%, 38.5%) p=0.0951
SDST: Avg Reaction Time (msec)	0.0196	-5.0 (-11.1, 1.2) p=0.1099	-6.0 (-12.1, 0.0) p=0.0501	1.2 (-4.9, 7.3) p=0.6968	-7.8 (-14.0, -1.6) p=0.0144	-2.2 (-6.6, 2.2) p=0.3231
SDST: # Correct Responses	0.6117	0.35 (-0.42, 1.12) p=0.3604	0.59 (-0.18, 1.35) p=0.1278	0.46 (-0.32, 1.24) p=0.2431	0.36 (-0.40, 1.12) p=0.3429	0.06 (-0.50, 0.63) p=0.8288
SDST: % Correct Responses (%)	0.3722	-0.4 (-1.2, 0.3) p=0.2173	-0.7 (-1.4, 0.0) p=0.0639	-0.5 (-1.2, 0.2) p=0.1893	-0.6 (-1.3, 0.1) p=0.1151	-0.0 (-0.6, 0.5) p=0.8982
SDST: # Incorrect Responses	0.0107	99.9 (-3.7, 203.4) p=0.0582	134.3 (32.3, 236.4) p=0.0114	9.3 (-94.2, 112.8) p=0.8561	149.7 (46.6, 252.7) p=0.0057	37.6 (-36.9, 112.1) p=0.3129
Adaptive Tracking (%)	0.0011	-0.721 (-3.514, 2.073) p=0.6055	-4.518 (-7.269, -1.767) p=0.0019	1.021 (-1.771, 3.812) p=0.4648	-3.221 (-5.996, -0.446) p=0.0240	-1.519 (-3.505, 0.467) p=0.1303
VAS Alertness (mm)	0.2004	2.1 (-0.7, 4.9) p=0.1448	-1.2 (-4.0, 1.6) p=0.3788	1.0 (-1.8, 3.8) p=0.4768	1.1 (-1.7, 3.9) p=0.4372	-0.6 (-2.7, 1.4) p=0.5399
VAS Calmness (mm)	0.0008	-0.3 (-3.5, 2.8) p=0.8264	-5.8 (-8.7, -2.8) p=0.0004	0.0 (-3.0, 3.0) p=0.9936	-3.6 (-6.5, -0.6) p=0.0206	-1.3 (-3.4, 0.9) p=0.2318

CONCLUSION

IV oliceridine has a reduced impact on several clinically relevant measures of cognitive performance, compared to IV morphine, including measures of sedation, motor performance, and eye-hand coordination.

REFERENCES

DeWire, S, Yamashita, DS, Rominger, DH, Liu, G, Cowan, CL, Graczyk, TM, Chen, X-T, Pitis, PM, Gotchev, D, Yuan, C, Koblish, M, Lark, MW, Violin, JD. *J Pharmacol Exp Ther 2013 Mar; 344:708–717.*

Liang, D-Y, Li, W-W, Nwaneshiudu, C, Irvine, K-A, Clark, JD. *Anesth Analg, 2019 Nov; 129(5):1414-1421.*

Muscata, SM, Deemsa, NP, D'Angelo, H, Kitt, MM, Gracee, PM, Andersen, ND, Silverman, SN, Rice, KC, Watkins, LR, Maier, SF, Barrientos, RM. *Neurobiology of Aging 2021; 98:214-224.*

Acknowledgments: We thank the subjects for their participation in this study.

Disclosure: Olinvyk® is manufactured by Trevena, Inc. and is approved by FDA for use in adults for the treatment of acute pain severe enough to require an opioid analgesic and for whom alternative treatments are inadequate. Full prescribing information is available at olinvyk.com.

Presented at the 64th annual meeting of The American College of Neuropsychopharmacology (ACNP), December 4-7. 2022