Preclinical dose scheduling studies of LOR-253, a novel anticancer drug, in combination with chemotherapeutics in lung and colon cancers

Howard Cukier, Robert Peralta, Hongnan Jin, Mario Huesca, Yoon Lee and Aiping Young

Lorus Therapeutics Inc. Toronto, Ontario, Canada

3710

Abstract

LOR-253 is an anticancer small molecule that is currently in a Phase I clinical study in patients with advanced or metastatic solid tumours. LOR-253 has a novel anticancer mechanism based on chelation of intracellular labile zinc, leading to inhibition of angiogenesis as well as G1/S cell cycle arrest due to induction of the tumour suppressor Kruppel-like factor 4 (KLF4). In preclinical studies, LOR-253 has shown potent antitumor activity against several cancers, including non-small cell lung cancer (NSCLC) and colon cancer, without significant toxicity. To support the clinical development of LOR-253 in combination with chemotherapeutics, we investigated the effect of dose scheduling of LOR-253 plus chemotherapy agents on anticancer activity in NSCLC (H226) and colon cancer (SW620) cell lines. Treatment of H226 cells in vitro with docetaxel, paclitaxel, or cisplatin for two days followed by increasing doses of LOR-253 resulted in significantly higher anti-proliferative activity than either agent alone. Assessment of drug interactions by determination of the combination index (D) (Berenbaum MC, Adv Cancer Res., 1981; 35:269-335) showed that the anticancer activities of the combination of LOR-253 with these chemotherapy agents were synergistic (D < 1). In studies with LOR-253 and docetaxel, synergistic anticancer activity against H226 was maintained with either concurrent or sequential treatments of each agent. Anticancer synergy was also seen in SW620 cells treated with LOR-253 in combination with oxaliplatin, CPT-11, or Fluorouracil. The effects of dose scheduling on activity of LOR-253 and docetaxel in vivo was also examined. Nude mice with H226 tumor xenografts were treated with repeat cycles of docetaxel once per week, followed two days later by treatment with LOR-253 for three consecutive days. Docetaxel was administered at an experimentally-determined ED_{50} dose level (defined as the effective dose at which 50% tumour inhibition was achieved), while LOR-253 was given at its maximum efficacious dose in this model (10 mg/kg) or sub-optimal dose (5 mg/kg). Sequential administration of docetaxel followed by LOR-253 showed significant antitumor activity when docetaxel was given at the ED_{50} dose, followed by 10 mg/kg or 5mg/kg of LOR-253, compared to either agent alone given at the same dose, or with concurrent administration of LOR-253 and docetaxel at these dose levels. Preliminary results also show significant antitumor activity of LOR-253 plus docetaxel against H226 tumours in mice treated with repeat cycles of the reverse order of these treatments (LOR-253 for three days, followed by docetaxel 1X/week). In summary, dose scheduling studies with LOR-253 plus chemotherapy drugs demonstrate strong anti-cancer activities in NSCLC and colon cancer, providing support for the design of LOR-253 combination strategies for treatment of these cancers.

In vitro sequential/concurrent treatment in H226 NSCLC cells

Cisplatin and LOR-253

Sequential: Cisplatin 2 days, then LOR-253 3 days	Result
1) Cisplatin IC50 then LOR-253 IC50	**Syn** D=0.51
2) Cisplatin IC50 then LOR-253 ½ IC50	**Syn** D=0.57
3) Cisplatin ½ IC50 then LOR-253 IC50	**Syn** D=0.80
4) Cisplatin ½ IC50 then LOR-253 ½ IC50	**Syn** D=0.76

Paclitaxel and LOR-253

Sequential: Paclitaxel 2 days, then LOR-253 3 days	Result	**Concurrent**: Paclitaxel + LOR-253 5 days	Result
1) Paclitaxel IC50 then LOR-253 IC50	**Syn** D=0.44	1) Paclitaxel IC50 + LOR-253 IC50	**Syn** D=0.43
2) Paclitaxel IC50 then LOR-253 ½ IC50	**Syn** D=0.30	2) Paclitaxel IC50 + LOR-253 ½ IC50	**Syn** D=0.30
3) Paclitaxel ½ IC50 then LOR-253 IC50	**Syn** D=0.42	3) Paclitaxel ½ IC50 + LOR-253 IC50	**Syn** D=0.42
4) Paclitaxel ½ IC50 then LOR-253 ½ IC50	**Syn** D=0.30	4) Paclitaxel ½ IC50 + LOR-253 ½ IC50	**Syn** D=0.30

Docetaxel and LOR-253

Reverse Sequential: LOR-253 3 days, then Docetaxel 2 days	Result	**Sequential**: Docetaxel 2 days, then LOR-253 3 days	Result	**Concurrent**: Docetaxel + LOR-253 over 5 days	Result
1) LOR-253 IC50 then Docetaxel IC50	**Syn** D=0.43	1) Docetaxel IC50 then LOR-253 IC50	**Syn** D=0.44	1) Docetaxel IC50 + LOR-253 IC50	**Syn** D=0.44
2) LOR-253 IC50 then Docetaxel ½ IC50	**Syn** D=0.45	2) Docetaxel IC50 then LOR-253 ½ IC50	**Syn** D=0.24	2) Docetaxel IC50 + LOR-253 ½ IC50	**Syn** D=0.26
3) LOR-253 ½ IC50 then Docetaxel IC50	**Syn** D=0.35	3) Docetaxel ½ IC50 then LOR-253 IC50	**Syn** D=0.43	3) Docetaxel ½ IC50 + LOR-253 IC50	**Syn** D=0.49
4) LOR-253 ½ IC50 then Docetaxel ½ IC50	**Syn** D=0.56	4) Docetaxel ½ IC50 then LOR-253 ½ IC50	**Syn** D=0.26	4) Docetaxel ½ IC50 + LOR-253 ½ IC50	**Syn** D=0.26

Result is based on formula:
Ac/Ae + Bc/Be = D (combination index)
Where: Ac and Bc correspond to the concentrations of drugs used in the combination treatment, and Ae and Be are the concentrations of drugs able to individually produce the same result.

D= <1 the effect of the combination is synergistic (Syn)
D= 1 the effect of the combination is additive (Add)
D= >1 if too high is antagonistic (Ant)

Figure 1. In vitro treatment of H226 cells with LOR-253 and chemotherapeutics. H226 cells were treated either sequentially or concurrently with LOR-253 as well as chemotherapeutics clinically relevant to NSCLC. For sequential and concurrent treatment, cells were treated with either the IC50 or ½ IC50 dose of chemotherapeutic along with increasing concentrations of LOR-253 (0.0026uM–50uM) for concurrent studies, or treated 48h later with increasing LOR-253 concentrations for sequential studies. After 5 days, cell proliferation analysis was performed. Reverse sequential studies were conducted similarly to sequential studies, but the order of treatment was reversed. Assessment of drug interactions was determined using the combination index (D) equation (Berenbaum MC, Adv Cancer Res., 1981; 35:269-335).

In vitro sequential/concurrent treatment in SW620 colon cancer cells

5-FU and LOR-253

Sequential: 5-FU 2 days, then LOR-253 3 days	Result	**Concurrent**: 5-FU + LOR-253, 5 days	Result
1) 5-FU ½ IC50 then LOR-253 IC50	**Add** D=1.06	1) 5-FU ½ IC50 + LOR-253 IC50	**Syn** D=0.50
2) 5-FU ½ IC50 then LOR-253 ½ IC50	**Ant** D=1.18	2) 5-FU ½ IC50 + LOR-253 ½ IC50	**Syn** D=0.94

CPT-11 and LOR-253

Concurrent: CPT-11 + LOR-253 over 5 days	Result
1) CPT-11 IC50 + LOR-253 IC50	**Add** D=1.15
2) CPT-11 IC50 + LOR-253 ½ IC50	**Add** D=1.04
3) CPT-11 ½ IC50 + LOR-253 IC50	**Syn** D=0.58
4) CPT-11 ½ IC50 + LOR-253 ½ IC50	**Syn** D=0.76

Oxaliplatin and LOR-253

Reverse Sequential: LOR-253 3 days, then Oxaliplatin 2 days	Result	**Sequential**: Oxaliplatin 2 days, then LOR-253 3 days	Result	**Concurrent**: Oxaliplatin + LOR-253 5 days	Result
1) LOR-253 IC50 then Oxaliplatin IC50	**Syn** D=0.48	1) Oxaliplatin IC50 then LOR-253 IC50	**Syn** D=0.56	1) Oxaliplatin IC50 + LOR-253 IC50	**Syn** D=0.48
2) LOR-253 IC50 then Oxaliplatin ½ IC50	**Syn** D=0.44	2) Oxaliplatin IC50 then LOR-253 ½ IC50	**Syn** D=0.35	2) Oxaliplatin IC50 + LOR-253 ½ IC50	**Syn** D=0.36
3) LOR-253 ½ IC50 then Oxaliplatin IC50	**Syn** D=0.27	3) Oxaliplatin ½ IC50 then LOR-253 IC50	**Syn** D=0.47	3) Oxaliplatin ½ IC50 + LOR-253 IC50	**Syn** D=0.44
4) LOR-253 ½ IC50 then Oxaliplatin ½ IC50	**Syn** D=0.24	4) Oxaliplatin ½ IC50 then + LOR-253 ½ IC50	**Syn** D=0.27	4) Oxaliplatin ½ IC50 + LOR-253 ½ IC50	**Syn** D=0.27

Figure 3. In vitro treatment of SW620 cells with LOR-253 and chemotherapeutics. SW620 cells were treated either sequentially or concurrently with LOR-253 as well as chemotherapeutics clinically relevant to colon cancer. For sequential and concurrent treatment, cells were treated with either the IC50 or ½ IC50 dose of chemotherapeutic along with increasing concentrations of LOR-253 (0.0026uM–50uM) for concurrent studies, or treated 48h later with increasing LOR-253 concentrations for sequential studies. After 5 days, cell proliferation analysis was performed. Concurrent treatment with ½ IC50 dose of 5-FU and CPT-11 and LOR-253 resulted in synergy, whereas the IC50 dose did not as it was not as effective in these studies.

In vitro sequential treatment

H226: LOR-253 and Docetaxel

SW620: LOR-253 and oxaliplatin

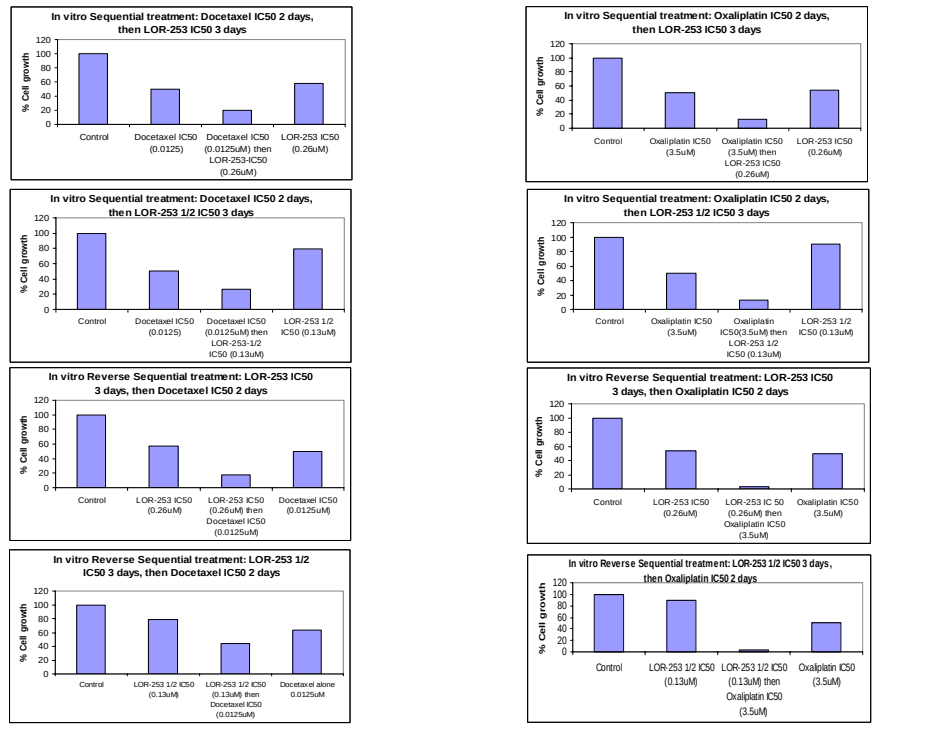
In vitro Sequential treatment: Docetaxel IC50 2 days, then LOR-253 IC50 3 days
In vitro Sequential treatment: Docetaxel IC50 2 days, then LOR-253 1/2 IC50 3 days
In vitro Reverse Sequential treatment: LOR-253 IC50 3 days, then Docetaxel IC50 2 days
In vitro Reverse Sequential treatment: LOR-253 1/2 IC50 3 days, then Docetaxel IC50 2 days
In vitro Sequential treatment: Oxaliplatin IC50 2 days, then LOR-253 IC50 3 days
In vitro Sequential treatment: Oxaliplatin IC50 2 days, then LOR-253 1/2 IC50 3 days
In vitro Reverse Sequential treatment: LOR-253 IC50 3 days, then Oxaliplatin IC50 2 days
In vitro Reverse Sequential treatment: LOR-253 1/2 IC50 3 days, then Oxaliplatin IC50 2 days
% Cell growth
Control

Figure 5. In vitro sequential treatment of H226 or SW620 cells. Analysis of in vitro sequential treatment results. H226 or SW620 cells were treated as indicated with IC50 or ½ IC50 levels of LOR-253, as well as, IC50 or ½ IC50 doses of docetaxel or oxaliplatin respectively. Sequential treatment involved treating cells for two days with chemotherapeutic at the indicated dose, followed by three days treatment with increasing concentrations of LOR-253. Reverse sequential treatment involved treating for three days with LOR-253, followed by two days treatment with increasing concentrations of the indicated chemotherapeutic.

Introduction

- LOR-253 is a second generation novel chemical entity currently in Phase I Clinical trial study in patients with advanced or metastatic solid tumours
- Treatment with LOR-253 mono-therapy in colon and NSCLC xenograft mouse models and cell lines demonstrates efficacy
- To support the clinical development of LOR-253 in combination with chemotherapeutics, we investigated the effect of dose scheduling of LOR-253 plus chemotherapy agents on anticancer activity in NSCLC (H226) and colon cancer (SW620) cell lines.
- Initial in vitro studies were conducted examining LOR-253 in combination with multiple chemotherapy agents clinically relevant to either NSCLC or colon cancer
- In vivo ED_{50} studies (defined as the effective dose at which 50% tumour inhibition was achieved) were carried out for the chemotherapeutic agents utilized in combination studies with LOR- 253
- LOR-253 and docetaxel were used in vivo in combination studies in an H226 xenograft mouse model and LOR-253 and oxaliplatin were used in vivo in combination studies in an SW620 xenograft mouse model

The present studies investigate the scheduling options for combination therapy involving LOR-253 plus chemotherapy on anti-cancer activity in NSCLC (H226) and colon cancer (SW620) cells, which will be used in the planning of future clinical studies

In vivo H226 xenograft combination studies

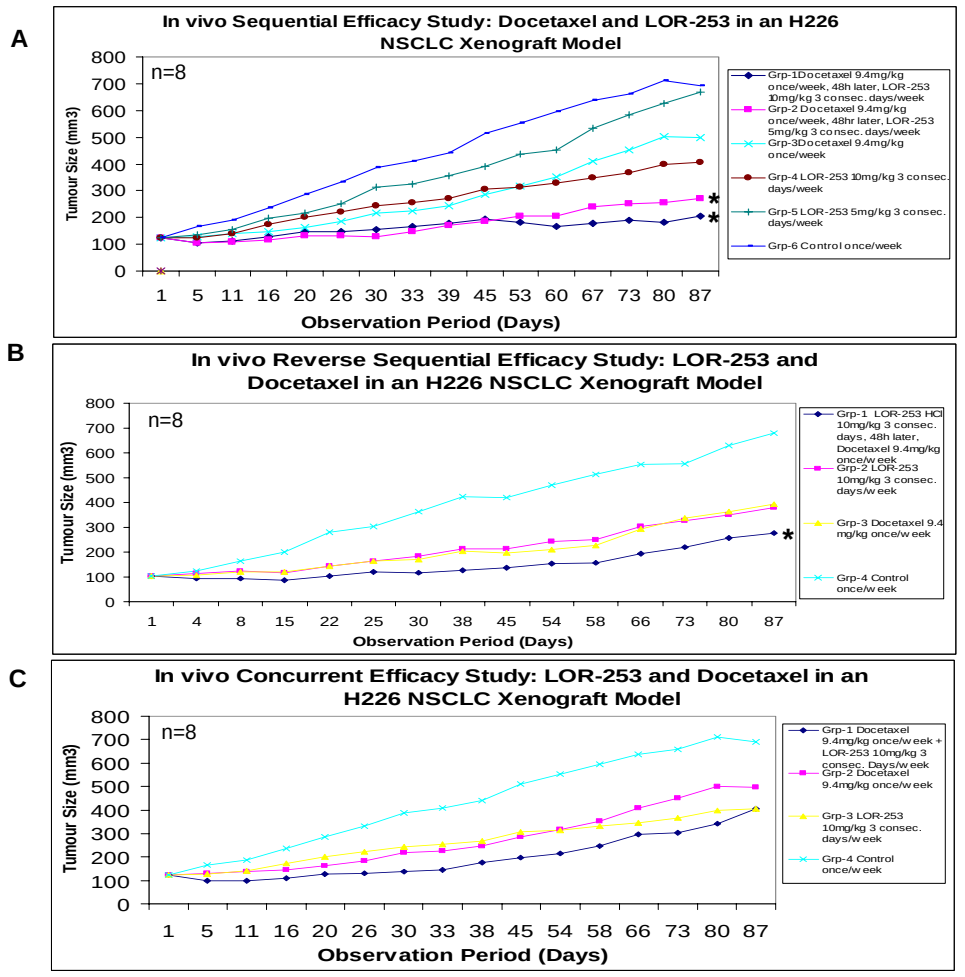
A
In vivo Sequential Efficacy Study: Docetaxel and LOR-253 in an H226 NSCLC Xenograft Model
n=8
Grp-1Docetaxel 9.4mg/kg once/week, 48h later, LOR-253 10mg/kg 3 consec. days/week
Grp-2 Docetaxel 9.4mg/kg once/week, 48hr later, LOR-253 5mg/kg 3 consec. days/week
Grp-3Docetaxel 9.4mg/kg once/week
Grp-4 LOR-253 10mg/kg 3 consec. days/week
Grp-5 LOR-253 5mg/kg 3 consec. days/week
Grp-6 Control once/week
B
In vivo Reverse Sequential Efficacy Study: LOR-253 and Docetaxel in an H226 NSCLC Xenograft Model
Grp-1 LOR-253 HCl 10mg/kg 3 consec. days, 48h later, Docetaxel 9.4mg/kg once/week
Grp-2 LOR-253 10mg/kg 3 consec. days/week
Grp-3 Docetaxel 9.4 mg/kg once/week
Grp-4 Control once/week
C
In vivo Concurrent Efficacy Study: LOR-253 and Docetaxel in an H226 NSCLC Xenograft Model
Grp-1 Docetaxel 9.4mg/kg once/week + LOR-253 10mg/kg 3 consec. Days/week
Grp-2 Docetaxel 9.4mg/kg once/week
Grp-3 LOR-253 10mg/kg 3 consec. days/week
Grp-4 Control once/week
Tumour Size (mm3)
Observation Period (Days)

Figure 2. In vivo combination studies testing a variety of dosing schedules in the H226 NSCLC xenograft model. A) Sequential treatment with docetaxel followed by LOR-253. Treatment with an ED_{50} (9.4mg/kg) dose of docetaxel 1x/week, followed 48h later by LOR-253 3x/week at an optimal dose (10mg/kg) or sub-optimal dose (5mg/kg) were conducted. Studies were conducted for four cycles and resulted in significant anti-cancer activity. **B)** Reverse sequential treatment, LOR-253 followed by docetaxel. Treatment with the optimal dose of LOR-253 3x/week, followed 24h later by ED_{50} dose of docetaxel 1x/week, were conducted over three cycles and resulted in significant anti-cancer activity. **C**) Concurrent treatment docetaxel plus LOR-253. Treatment with ED_{50} dose of docetaxel 1x/week, along with the optimal dose of LOR-253 3x/week, over four cycles did not result in significant anti-cancer activity. ED_{50} studies with docetaxel and optimal dose testing with LOR-253 were determined previously. * P<0.05

In vivo SW620 xenograft combination studies

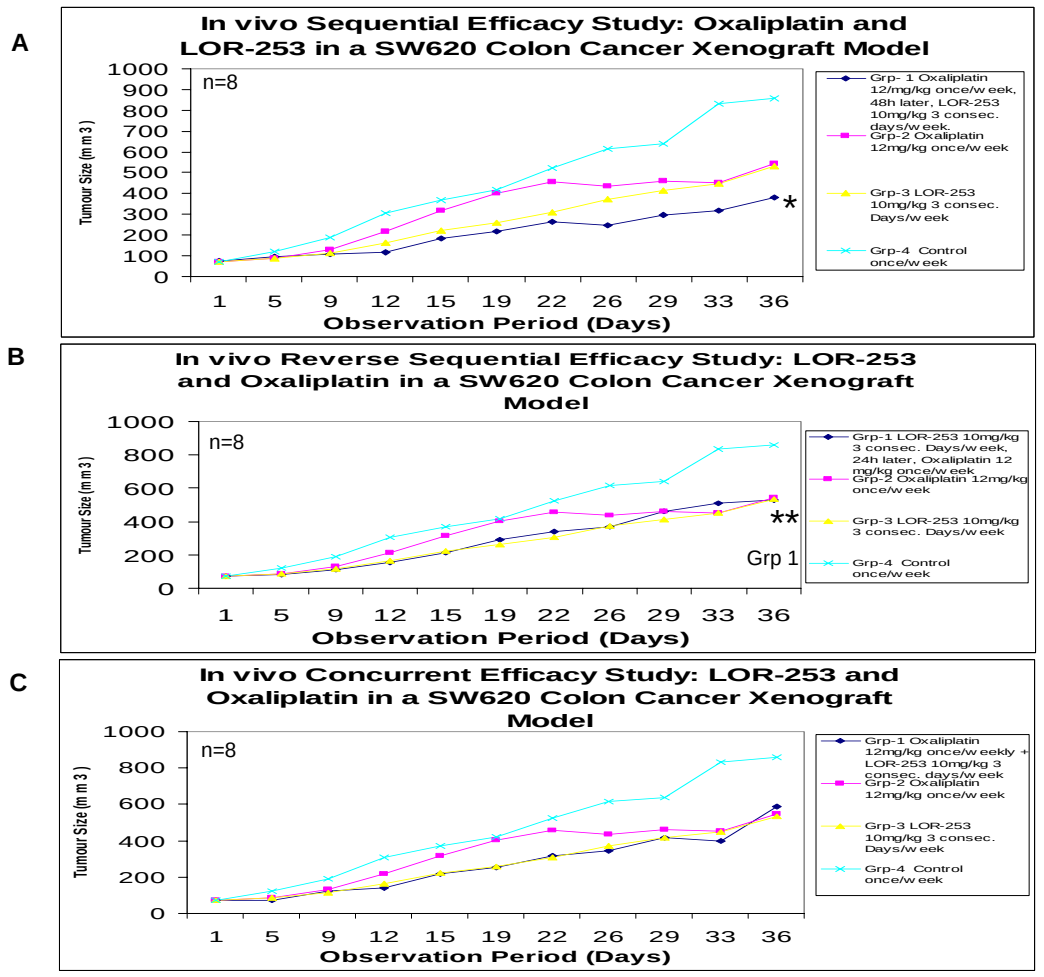
A
In vivo Sequential Efficacy Study: Oxaliplatin and LOR-253 in a SW620 Colon Cancer Xenograft Model
n=8
Grp- 1 Oxaliplatin 12/mg/kg once/week, 48h later, LOR-253 10mg/kg 3 consec. days/week.
Grp-2 Oxaliplatin 12mg/kg once/week
Grp-3 LOR-253 10mg/kg 3 consec. Days/week
Grp-4 Control once/week
B
In vivo Reverse Sequential Efficacy Study: LOR-253 and Oxaliplatin in a SW620 Colon Cancer Xenograft Model
Grp 1
Grp-1 LOR-253 10mg/kg 3 consec. Days/week, 24h later, Oxaliplatin 12 mg/kg once/week
Grp-2 Oxaliplatin 12mg/kg once/week
Grp-3 LOR-253 10mg/kg 3 consec. Days/week
Grp-4 Control once/week
C
In vivo Concurrent Efficacy Study: LOR-253 and Oxaliplatin in a SW620 Colon Cancer Xenograft Model
Grp-1 Oxaliplatin 12mg/kg once/weekly + LOR-253 10mg/kg 3 consec. days/week
Grp-2 Oxaliplatin 12mg/kg once/week
Grp-3 LOR-253 10mg/kg 3 consec. Days/week
Grp-4 Control once/week
Tumour Size (mm3)
Observation Period (Days)

Figure 4. In vivo combination studies testing a variety of dosing schedules in the SW620 colon cancer xenograft model. A) Sequential treatment with oxaliplatin followed by LOR-253. Treatment with a sub-ED_{50} (12mg/kg) dose of oxaliplatin 1x/week, followed 48h later by LOR-253 3x/week at an optimal dose (10mg/kg) were conducted. Studies were conducted for three cycles and resulted in significant anti-cancer activity. **B)** Reverse sequential treatment, LOR-253 followed by oxaliplatin. Treatment with the optimal dose of LOR-253 3x/week, followed 24h later by sub-ED_{50} dose of oxaliplatin 1x/week, were conducted over three cycles and resulted in significant anti-cancer activity. **C**) Concurrent treatment oxaliplatin plus LOR-253. Treatment with sub-ED_{50} dose of oxaliplatin 1x/week, along with the optimal dose of LOR-253 3x/week, over three cycles did not result in significant anti-cancer activity. ED_{50} studies with oxaliplatin and optimal dose testing with LOR-253 were determined previously. * P<0.05; ** P=0.05

Summary of Results

- Synergy is observed when LOR-253 is used in sequential treatment with docetaxel, paclitaxel or cisplatin and in concurrent treatment with paclitaxel or docetaxel, in H226 cells in vitro
- Synergy is also observed when LOR-253 is used in sequential or concurrent treatment with oxaliplatin and in concurrent treatment with 5-FU or CPT-11, in SW620 cells in vitro
- Sequential treatment with a sub-ED_{50} dose of oxaliplatin followed by the optimal dose of LOR-253 in a SW620 xenograft model resulted in significant anti-cancer activity; Significant results were also observed when the reverse sequence of treatment was administered
- Significant anti-cancer activity was observed in an H226 xenograft model when treated sequentially with the ED_{50} dose of docetaxel, followed by the optimal or sub-optimal dose of LOR-253; Significant results were also observed when the reverse sequence of treatment was administered
- In summary, dose scheduling studies with LOR-253 plus chemotherapy drugs demonstrate strong anti-cancer activities in NSCLC and colon cancer, providing support for the design of LOR-253 combination strategies for treatment of these cancers in future clinical studies

Methods

Cell proliferation studies

96 well plates were seeded with H226 or SW620 cells. The following day media was removed from the plates and compounds diluted in media were added in triplicate. For sequential studies, at the specified time point, media was removed, plates washed with PBS, and the subsequent compound was diluted in media and added to the cells. For concurrent studies, both compounds were added at the same time.

In vitro cell growth inhibition assay

After treatment as indicated at 37ºC for 5 days, cell viability was quantitated using the XTT (sodium 3'-[1-(phenylamino-carbonyl)-3,4-tetrazolium}-bis (4-methoxy-6-nitro) benzene sulfonic acid hydrate) colorimetric assay following manufacturers protocol (Roche Applied Science).

Mouse xenograft model

CD1 athymic nude mice (Charles River) were injected subcutaneously in the right flank region with 5 x 10^6 human H226 NSCLC or SW620 colon cancer cells. When tumours reached the desired size (100-120mm^3) studies were initiated. All drugs were administered intravenously according the specified treatment schedule, at the indicated doses. All animal studies were reviewed and approved by the animal care committee at Lorus Therapeutics.