Targeting Myeloid-Derived Suppressor Cells with Actinium-225 Lintuzumab, a CD33 Antibody Radioconjugate to Enhance Antitumor Immunity

Amanda Chin, Mary Chen, Jason Li, Debbie Lewis, Megan McCloskey, Caroline Jennings, Le-Cun Xu, Monideepa Roy, Denis Beckford-Vera and Helen Kotanides

Actinium Pharmaceuticals, New York, NY USA

Abstract # 4421

BACKGROUND

Myeloid-derived suppressor cells (MDSCs) are a heterogeneous population of myeloid lineage cells with potent immunosuppressive activity found enriched in cancer patients. Crosstalk between MDSCs and the tumor microenvironment can promote tumor immune evasion and thus there is growing interest in developing therapeutic approaches to intervene in the MDSC suppressive function. Both monocytic-MDSC (M-MDSC) and granulocytic-MDSC (G-MDSC) subpopulations express CD33 surface molecule. For well-defined cell surface markers like CD33, there is considerable interest in the use of radionuclides as therapeutic payloads, particularly α-particle emitters such as actinium-225 (^{225}Ac) since they deliver substantially higher decay energies over a much shorter distance than β-emitters, rendering them more suitable for precise, potent, and efficient target cell killing while minimizing toxicity to surrounding bystander cells. Actimab-A, the anti-CD33 antibody lintuzumab armed with the ^{225}Ac radioisotope (^{225}Ac-lintuzumab, CD33 ARC), is currently being evaluated in relapsed/refractory AML and has demonstrated significant anti-leukemic activity in Phase 1/2 clinical trials. We therefore hypothesized that MDSCs can be directly targeted by the CD33 ARC. Hence, we evaluated the therapeutic potential of the CD33 ARC to deplete MDSCs through preclinical studies in vitro and in vivo with a humanized mouse model.

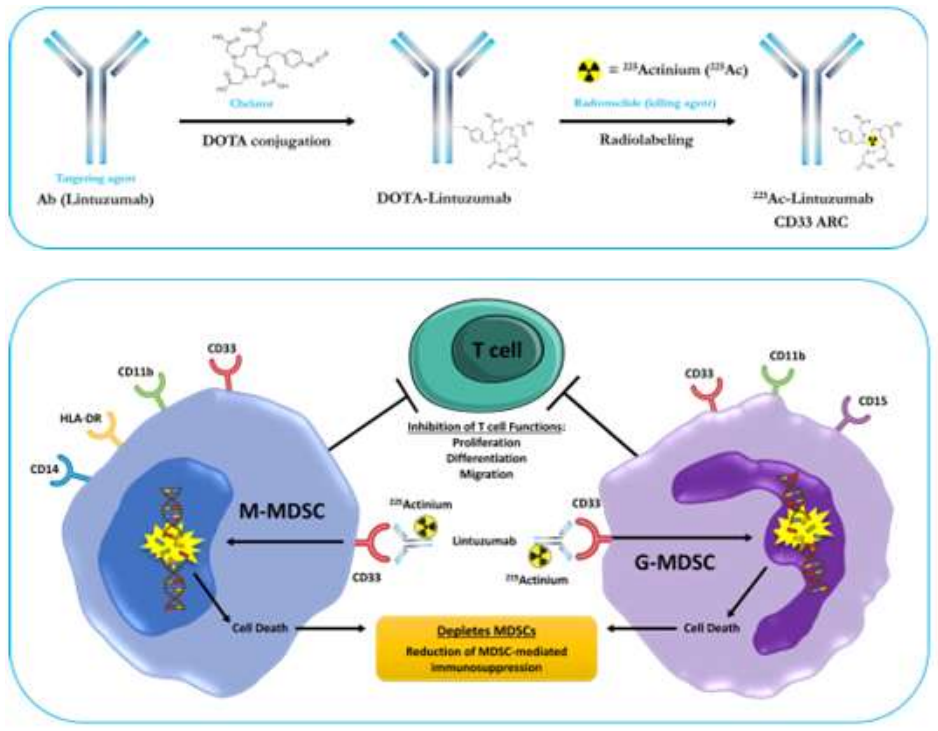
= ^{225}Actinium (^{225}Ac)
DOTA conjugation
Radiolabeling
Ab (Lintuzumab)
DOTA-Lintuzumab
^{225}Ac-Lintuzumab
CD33 ARC
T cell
Inhibition of T cell Functions:
Proliferation
Differentiation
Migration
M-MDSC
G-MDSC
CD11b
CD33
HLA-DR
CD14
CD15
Lintuzumab
Cell Death
Depletes MDSCs
Reduction of MDSC-mediated immunosuppression

Figure 1. **(A)** Schematic illustrating the preparation of CD33 antibody radioconjugate (ARC) armed with Actinium-225 (^{225}Ac). **(B)** Model showing mechanism of CD33 ARC therapy mediated targeting of MDSCs to enhance antitumor immunity.

METHODS

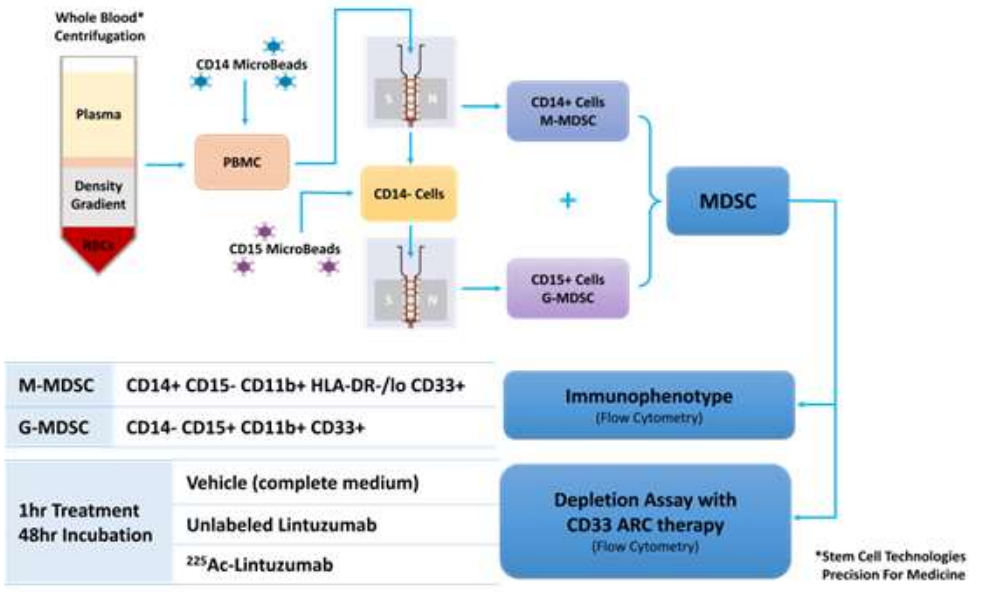
Whole Blood* Centrifugation
Plasma
Density Gradient
RBCs
PBMC
CD14 MicroBeads
CD15 MicroBeads
CD14- Cells
CD14+ Cells M-MDSC
CD15+ Cells G-MDSC
MDSC
M-MDSC CD14+ CD15- CD11b+ HLA-DR-/lo CD33+
G-MDSC CD14- CD15+ CD11b+ CD33+
Immunophenotype (Flow Cytometry)
1hr Treatment 48hr Incubation
Vehicle (complete medium)
Unlabeled Lintuzumab
225Ac-Lintuzumab
Depletion Assay with CD33 ARC therapy (Flow Cytometry)
*Stem Cell Technologies Precision For Medicine

Figure 2. Schematic representation of MDSC isolation from patient peripheral blood and subsequent analysis methods.

RESULTS

MDSC Characterization

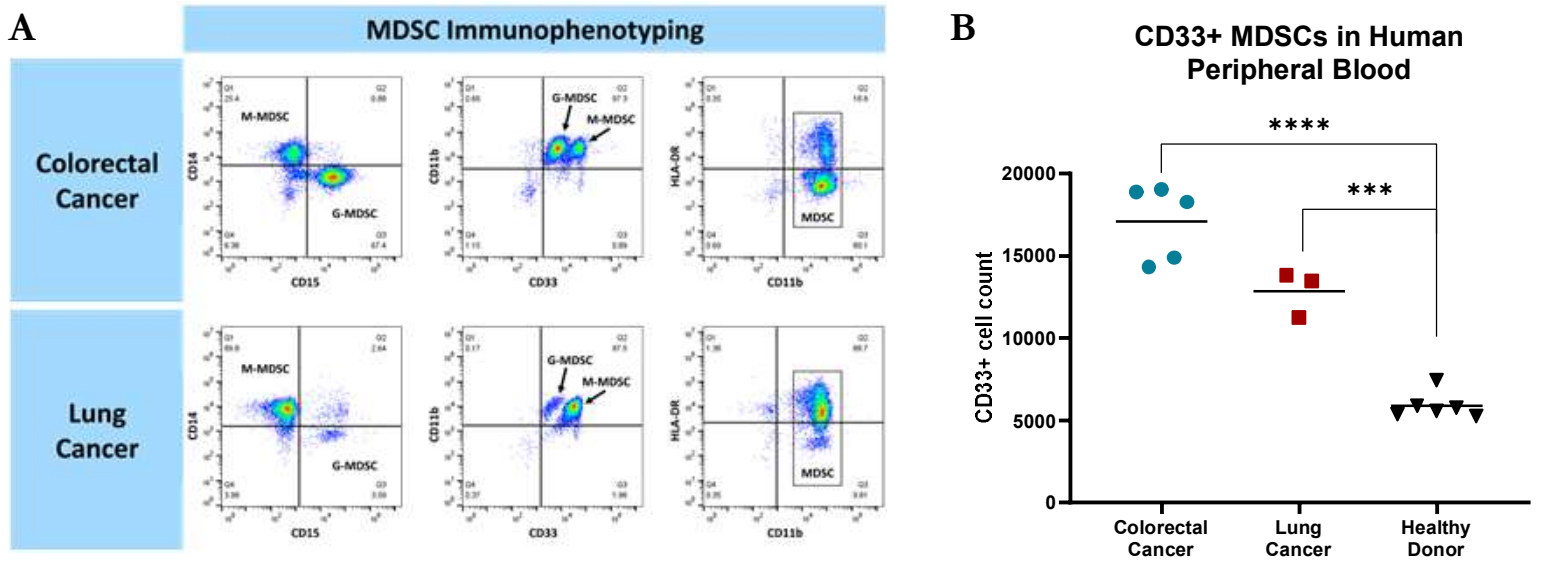
A
MDSC Immunophenotyping
Colorectal Cancer
Lung Cancer
M-MDSC
G-MDSC
MDSC
CD14
CD15
CD11b
CD33
HLA-DR
B
CD33+ MDSCs in Human Peripheral Blood
CD33+ cell count

Colorectal Cancer
Lung Cancer
Healthy Donor

Figure 3. CD33+ human MDSC isolation from the peripheral blood of cancer patients and healthy donors. **(A)** Flow cytometry immunophenotyping of MDSC populations isolated from human blood. Representative examples shown from a colorectal and a lung cancer patient. Granulocytic-MDSC (G-MDSC) are defined as CD14-CD15+CD11b+CD33+. Monocytic-MDSC (M-MDSC) are defined as CD14+CD15-CD11b+CD33+HLA-DR-/lo. G-MDSC display a lower CD33 expression in comparison to M-MDSC. **(B)** MDSCs, CD33+CD14+ or CD33+CD15+, are enriched in the blood of colorectal cancer patients (n=5, blue circle) and lung cancer patients (n=3, red square) in comparison to that of healthy donors (n=6, black triangles). Statistical analysis: t test (GraphPad Prism) ***$p<0.0005$, ****$p<0.0001$

CD33 ARC Treatment Decreases MDSC Viability Ex-Vivo

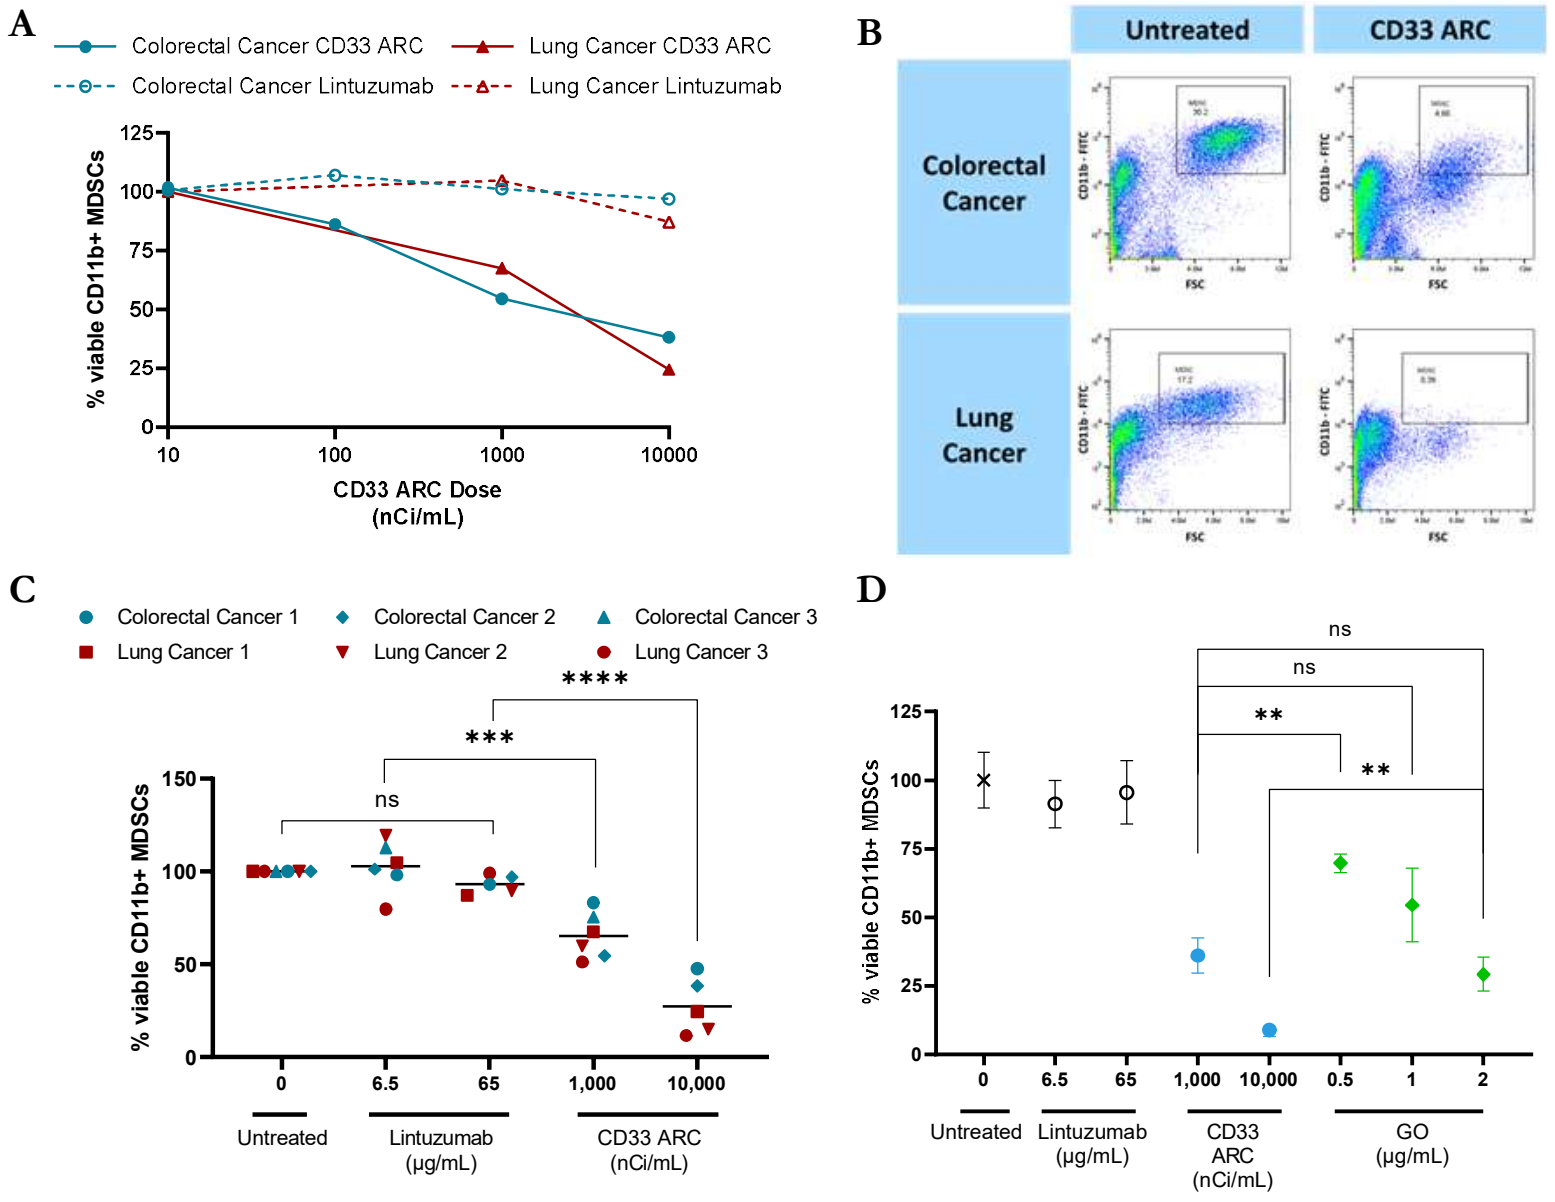
A
Colorectal Cancer CD33 ARC
Lung Cancer CD33 ARC
Colorectal Cancer Lintuzumab
Lung Cancer Lintuzumab
% viable CD11b+ MDSCs
CD33 ARC Dose (nCi/mL)
B
Untreated
CD33 ARC
Colorectal Cancer
Lung Cancer
C
Colorectal Cancer 1
Colorectal Cancer 2
Colorectal Cancer 3
Lung Cancer 1
Lung Cancer 2
Lung Cancer 3
Untreated
Lintuzumab (µg/mL)
CD33 ARC (nCi/mL)
D
Untreated
Lintuzumab (µg/mL)
CD33 ARC (nCi/mL)
GO (µg/mL)

Figure 4. CD33 ARC dose dependent depletion of human MDSCs derived from colorectal and lung cancer patient peripheral blood. **(A)** Human CD14+ and CD15+ MDSCs were treated with CD33 ARC, unlabeled (cold) lintuzumab antibody as control or left untreated for 48 hours. MDSC viability was measured using CD11b as cell surface marker and calculated as percent viable treated cells relative to untreated cells. Treatment with CD33 ARC resulted in a dose dependent decrease in MDSC viability in both colorectal and lung cancer MDSC samples. **(B)** Representative flow cytometry histograms demonstrate prominent CD11b+ MDSC population in untreated cells (left column) while significantly reducing this population in response to 10,000 nCi/mL CD33 ARC treatment (right column). **(C)** MDSC depletion ex-vivo was observed across multiple cancer patient samples (n=3 each for colorectal and lung) in response to CD33 ARC. Lintuzumab treatments exhibited no significant change to cell viability. **(D)** Colorectal cancer blood MDSC treated with CD33 ARC (blue circle) at 10,000 nCi/mL dose demonstrated more potent MDSC depletion relative to gemtuzumab ozogamicin (GO, Mylotarg; green diamond). GO dosing selected based on the human PK (C_{max}=2.86 mg/L for 9 mg/m^2 dose). For **A)** to **D)** unlabeled lintuzumab was used at mass equivalent concentrations to the CD33 ARC treatments. Statistical analysis: t test (GraphPad Prism) ns=not significant, **$p<0.01$, ***$p<0.0005$, ****$p<0.0001$

CD33 ARC Depletes MDSCs in a Humanized Mouse Model In Vivo

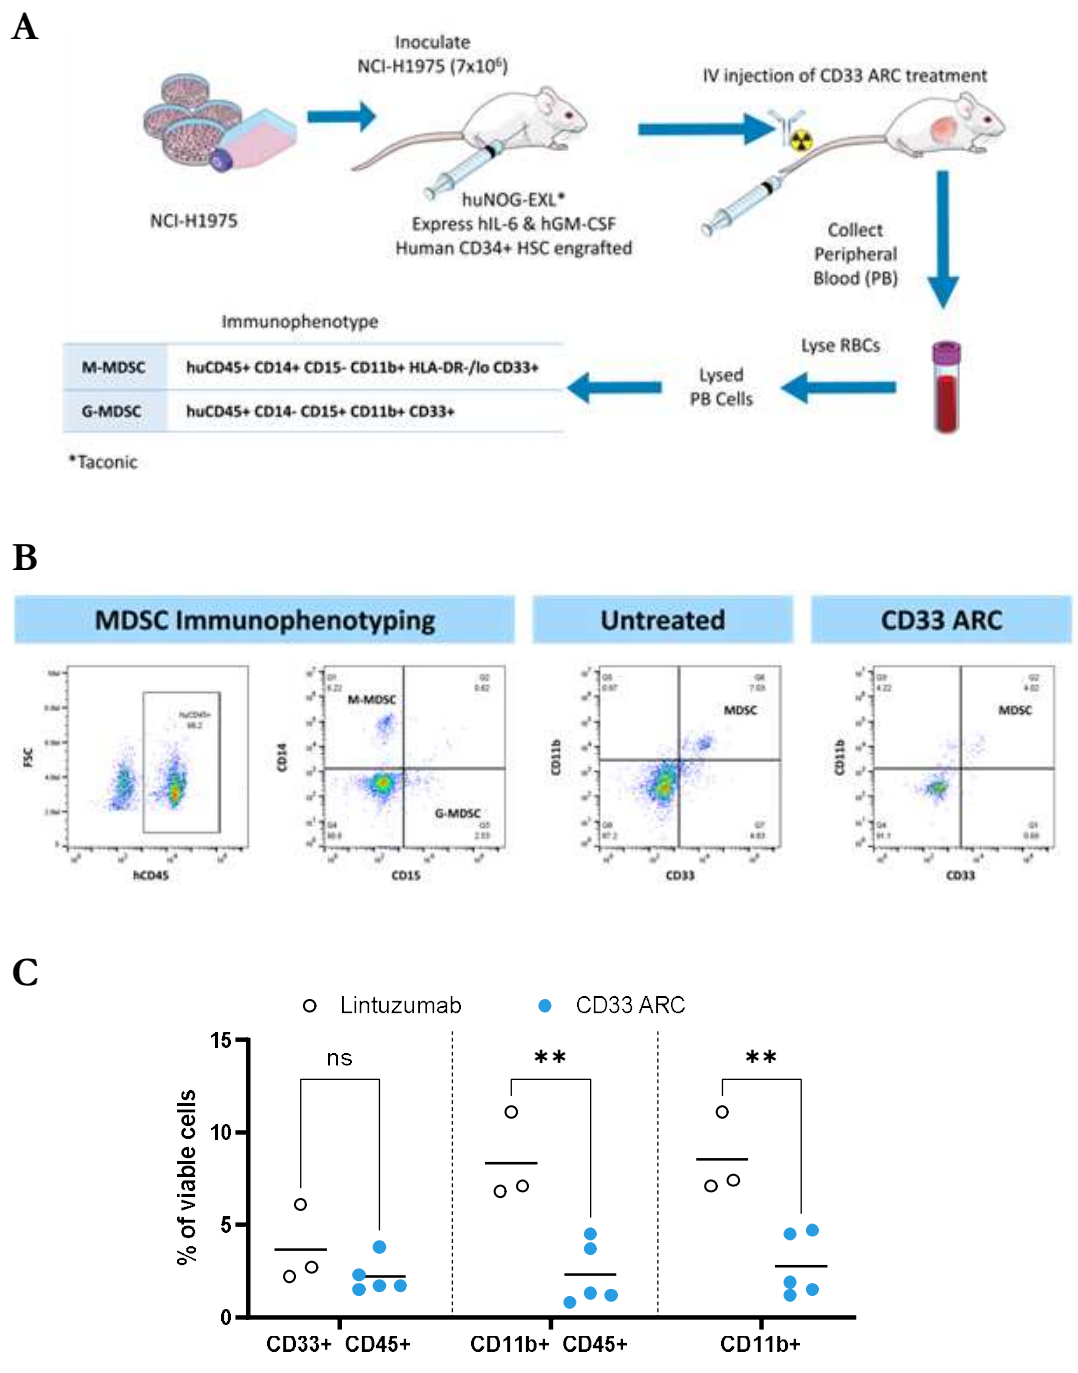
A
Inoculate NCI-H1975 (7x10^6)
IV injection of CD33 ARC treatment
NCI-H1975
huNOG-EXL*
Express hIL-6 & hGM-CSF
Human CD34+ HSC engrafted
Collect Peripheral Blood (PB)
Lyse RBCs
Lysed PB Cells
Immunophenotype
M-MDSC huCD45+ CD14+ CD15- CD11b+ HLA-DR-/lo CD33+
G-MDSC huCD45+ CD14- CD15+ CD11b+ CD33+
*Taconic
B
MDSC Immunophenotyping
Untreated
CD33 ARC
C
Lintuzumab
CD33 ARC
% of viable cells
CD33+ CD45+
CD11b+ CD45+
CD11b+

Figure 5. CD33 ARC targeted depletion of human MDSC in vivo. **(A)** Human CD34+ HSC engrafted NOG-EXL mice (Taconic) were treated intravenously with 80nCi CD33 ARC, unlabeled lintuzumab or untreated controls. After 6 or 10 days, peripheral blood samples were collected, RBCs lysed and MDSC populations immunophenotyped by flow cytometry. **(B)** Representative histograms of human MDSC population in blood of huNOG-EXL mice. Prior to treatment huCD45+CD14+CD33+CD11b+ and huCD45+CD15+CD33+CD11b+ MDSCs were detected. After 6 days of CD33 ARC treatment, these MDSC populations were depleted compared to untreated mice. **(C)** Flow cytometry viability analysis of blood MDSCs in tumor bearing huNOG-EXL mice treated for 10 days with CD33 ARC or unlabeled lintuzumab. CD33 ARC treatment significantly decreased the percentage of viable CD11b+CD45+ cells compared to unlabeled lintuzumab. Statistical analysis: t test (GraphPad Prism) ns=not significant, **$p<0.01$

CONCLUSIONS

- **CD33 ARC ^{225}Ac-Lintuzumab mediated the targeted cell death of human MDSCs ex vivo across cancer patient samples**
- **CD33 ARC treatment of NOG-EXL humanized mice in vivo significantly depleted human blood MDSCs and merits the evaluation of intratumor MDSCs in this model**
- **In summary, these preclinical findings suggest the potential to utilize CD33 ARC alpha targeted radiotherapy as an MDSC depleting agent to reduce immunosuppressive functions of MDSC and thereby enhance antitumor immunity which warrants further evaluation in cancer patients**

American Association for Cancer Research (AACR) Annual Meeting. Orlando, FL April 14-19, 2023