T cell infiltration into tumors induced by DPX-Survivac combination immunotherapy demonstrated by PET/MR imaging in an orthotopic ovarian cancer model

Poster# 384

AACR Virtual Meeting II 2020

Caitrin Sobey-Skelton[2], Marie-Laurence Tremblay[1], Hailey Wyatt[2], Victoria Gonzalez[2], Andrea Nuschke[3,4], Christa Davis[4], Alecia Mackay[1], Kim Bobbit[1], Andrea West[2], Barbara Vanderhyden[5], Genevieve Weir[6], Alexandra Merkx-Jacques[1], Kimberly Brewer[2,4], Marianne Stanford[1]

1. IMV Inc, Dartmouth, NS, Canada; 2. Dalhousie University, Halifax, NS, Canada; 3. Carleton University, Ottawa, ON, Canada; 4. BIOTIC, IWK Health Centre, Halifax, NS, Canada; 5. Ottawa Hospital Research Institute, Ottawa, ON, Canada; 6. Avanda BioLabs, Halifax, NS, Canada. Contact: amerkx-jacques@imv-inc.com

ABSTRACT & BACKGROUND

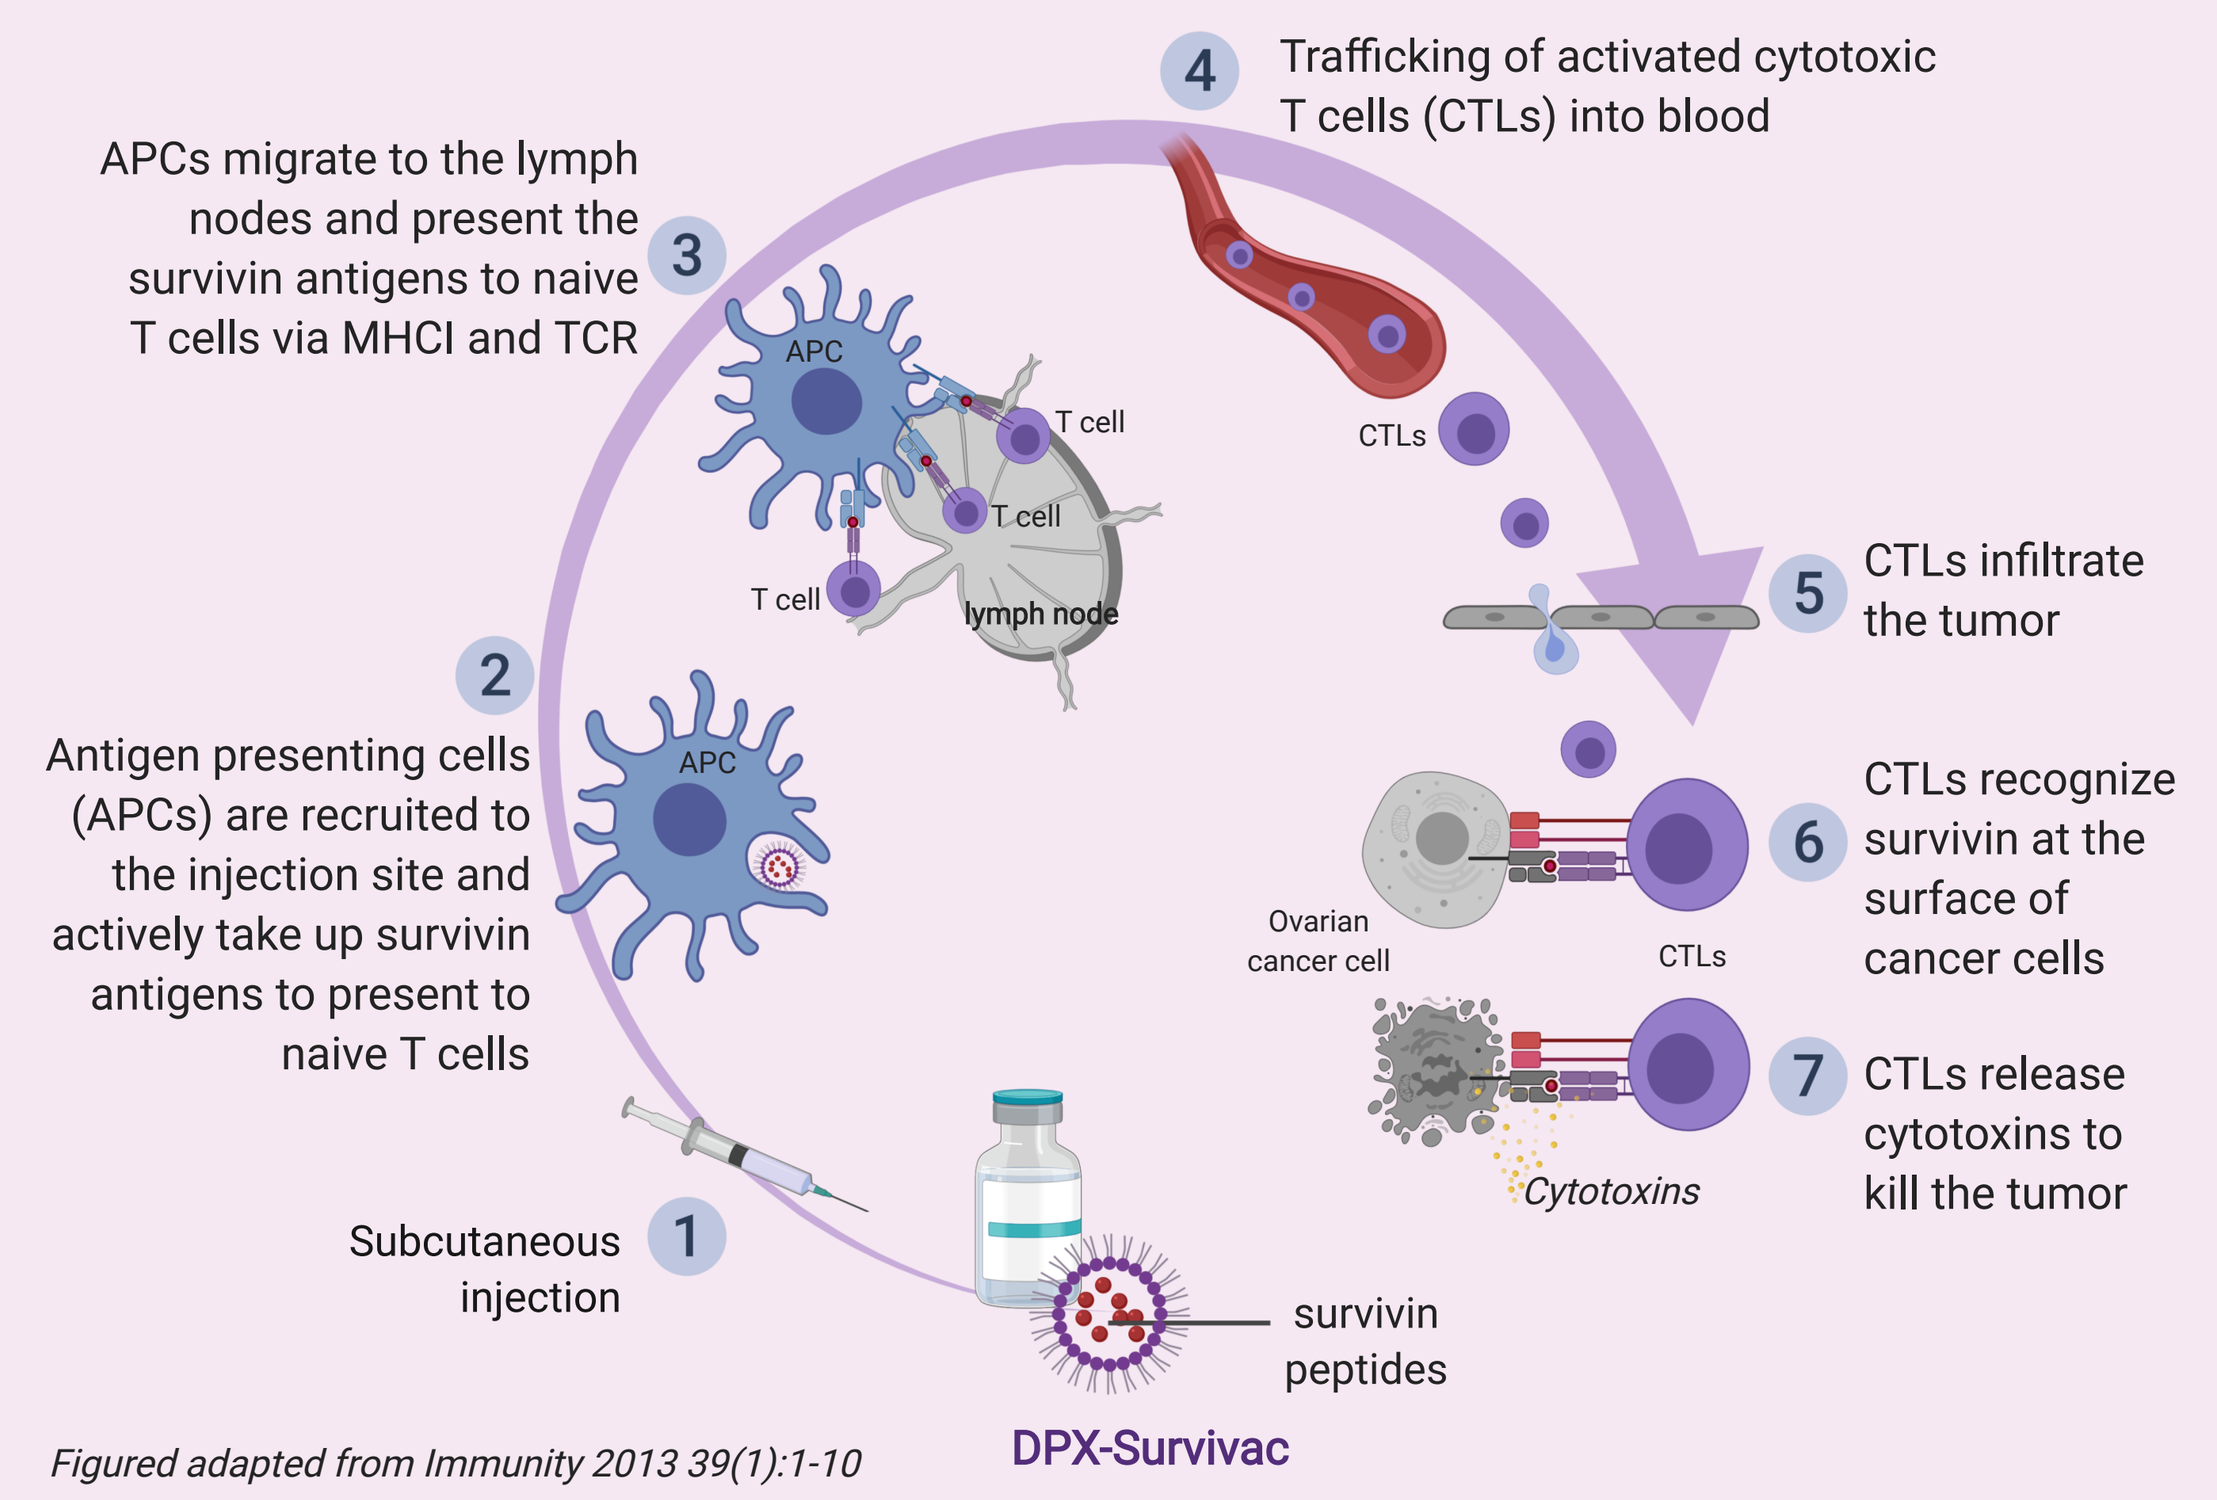
Trafficking of activated cytotoxic T cells (CTLs) into blood
APCs migrate to the lymph nodes and present the survivin antigens to naive T cells via MHCI and TCR
CTLs infiltrate the tumor
Antigen presenting cells (APCs) are recruited to the injection site and actively take up survivin antigens to present to naive T cells
CTLs recognize survivin at the surface of cancer cells
CTLs release cytotoxins to kill the tumor
Subcutaneous injection
survivin peptides
DPX-Survivac

Figured adapted from Immunity 2013 39(1):1-10

Epithelial ovarian cancers are highly aggressive and often diagnosed at late stages. To date, a limited number of patients benefit from immunotherapies, and recurrent ovarian cancer remains an area of unmet medical need. Novel immunotherapies, such as DPX-Survivac, that generate and drive activated T cells into tumors represent a promising approach. **To better understand DPX-Survivac's mechanism of action in ovarian cancer, we tracked immune cells using molecular imaging and studied immune infiltration with biological assays.**

OBJECTIVES

In a preclinical, orthotopic ovarian model:

* Compare tumor volumes of untreated mice to mice treated with a combination of DPX-Survivac with cyclophosphamide (CPA) and anti-PD-1 using positron emission tomography/magnetic resonance imaging (PET/MRI);
* Evaluate recruitment of $CD8^+$ cytotoxic T lymphocytes (CTLs) and myeloid cells (MCs) to tumors and lymph nodes *in vivo* using quantitative MRI cell tracking;
* Evaluate immune infiltration in ascites and in tumors *ex vivo*

METHODS

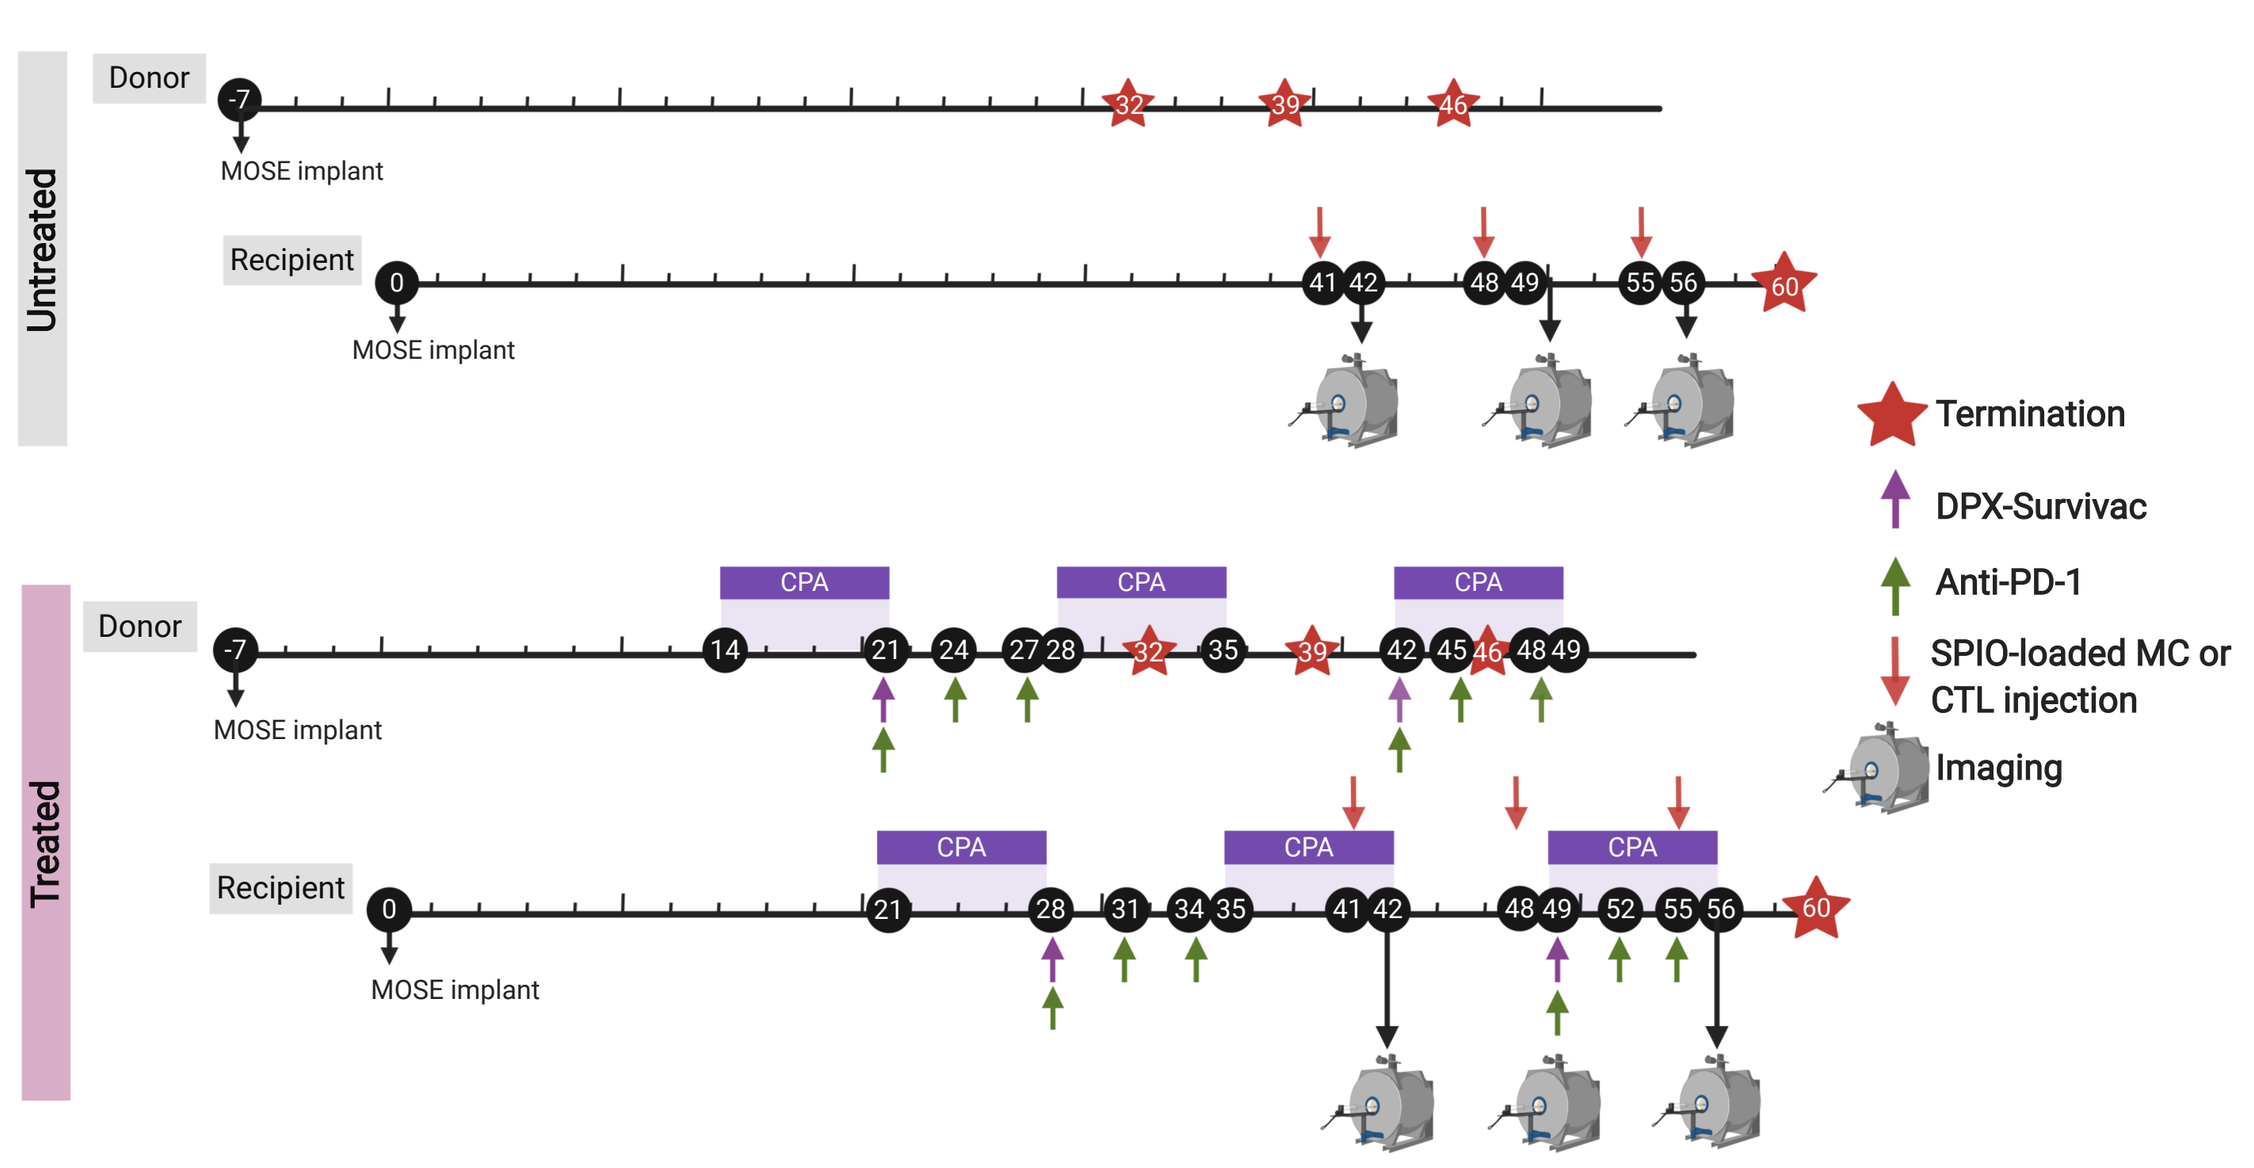
Untreated
Treated
Donor
Recipient
MOSE implant
CPA
Termination
DPX-Survivac
Anti-PD-1
SPIO-loaded MC or CTL injection
Imaging

Humanized transgenic mice (HLA-A2.1/HLA-DR1; H-2 class-I/II knockout) were implanted with 10^4 syngeneic mouse ovarian surface epithelial (MOSE) cells in the left ovarian bursa and were either untreated or treated with a combination of DPX-Survivac, CPA and anti-PD-1. The $CD8^+$ CTLs and MCs were isolated from disease matched donor mice, cultured *in vitro*, loaded with superparamagnetic iron oxide (SPIO) nanoparticles, and i.v. injected in recipient mice. The next day, recipient mice were injected with 500µCi of ^{18}F-fluorodeoxyglucose and imaged by MRI and PET. Anatomical images and tumor volumes were collected with a bSSFP, cells were semi-quantified using R_2* maps from a multi-echo single point imaging sequence (TurboSPI). Tumor metabolism was assessed by simultaneous acquisition of PET.

RESULTS

PET/MRI Analysis and Tumor Growth

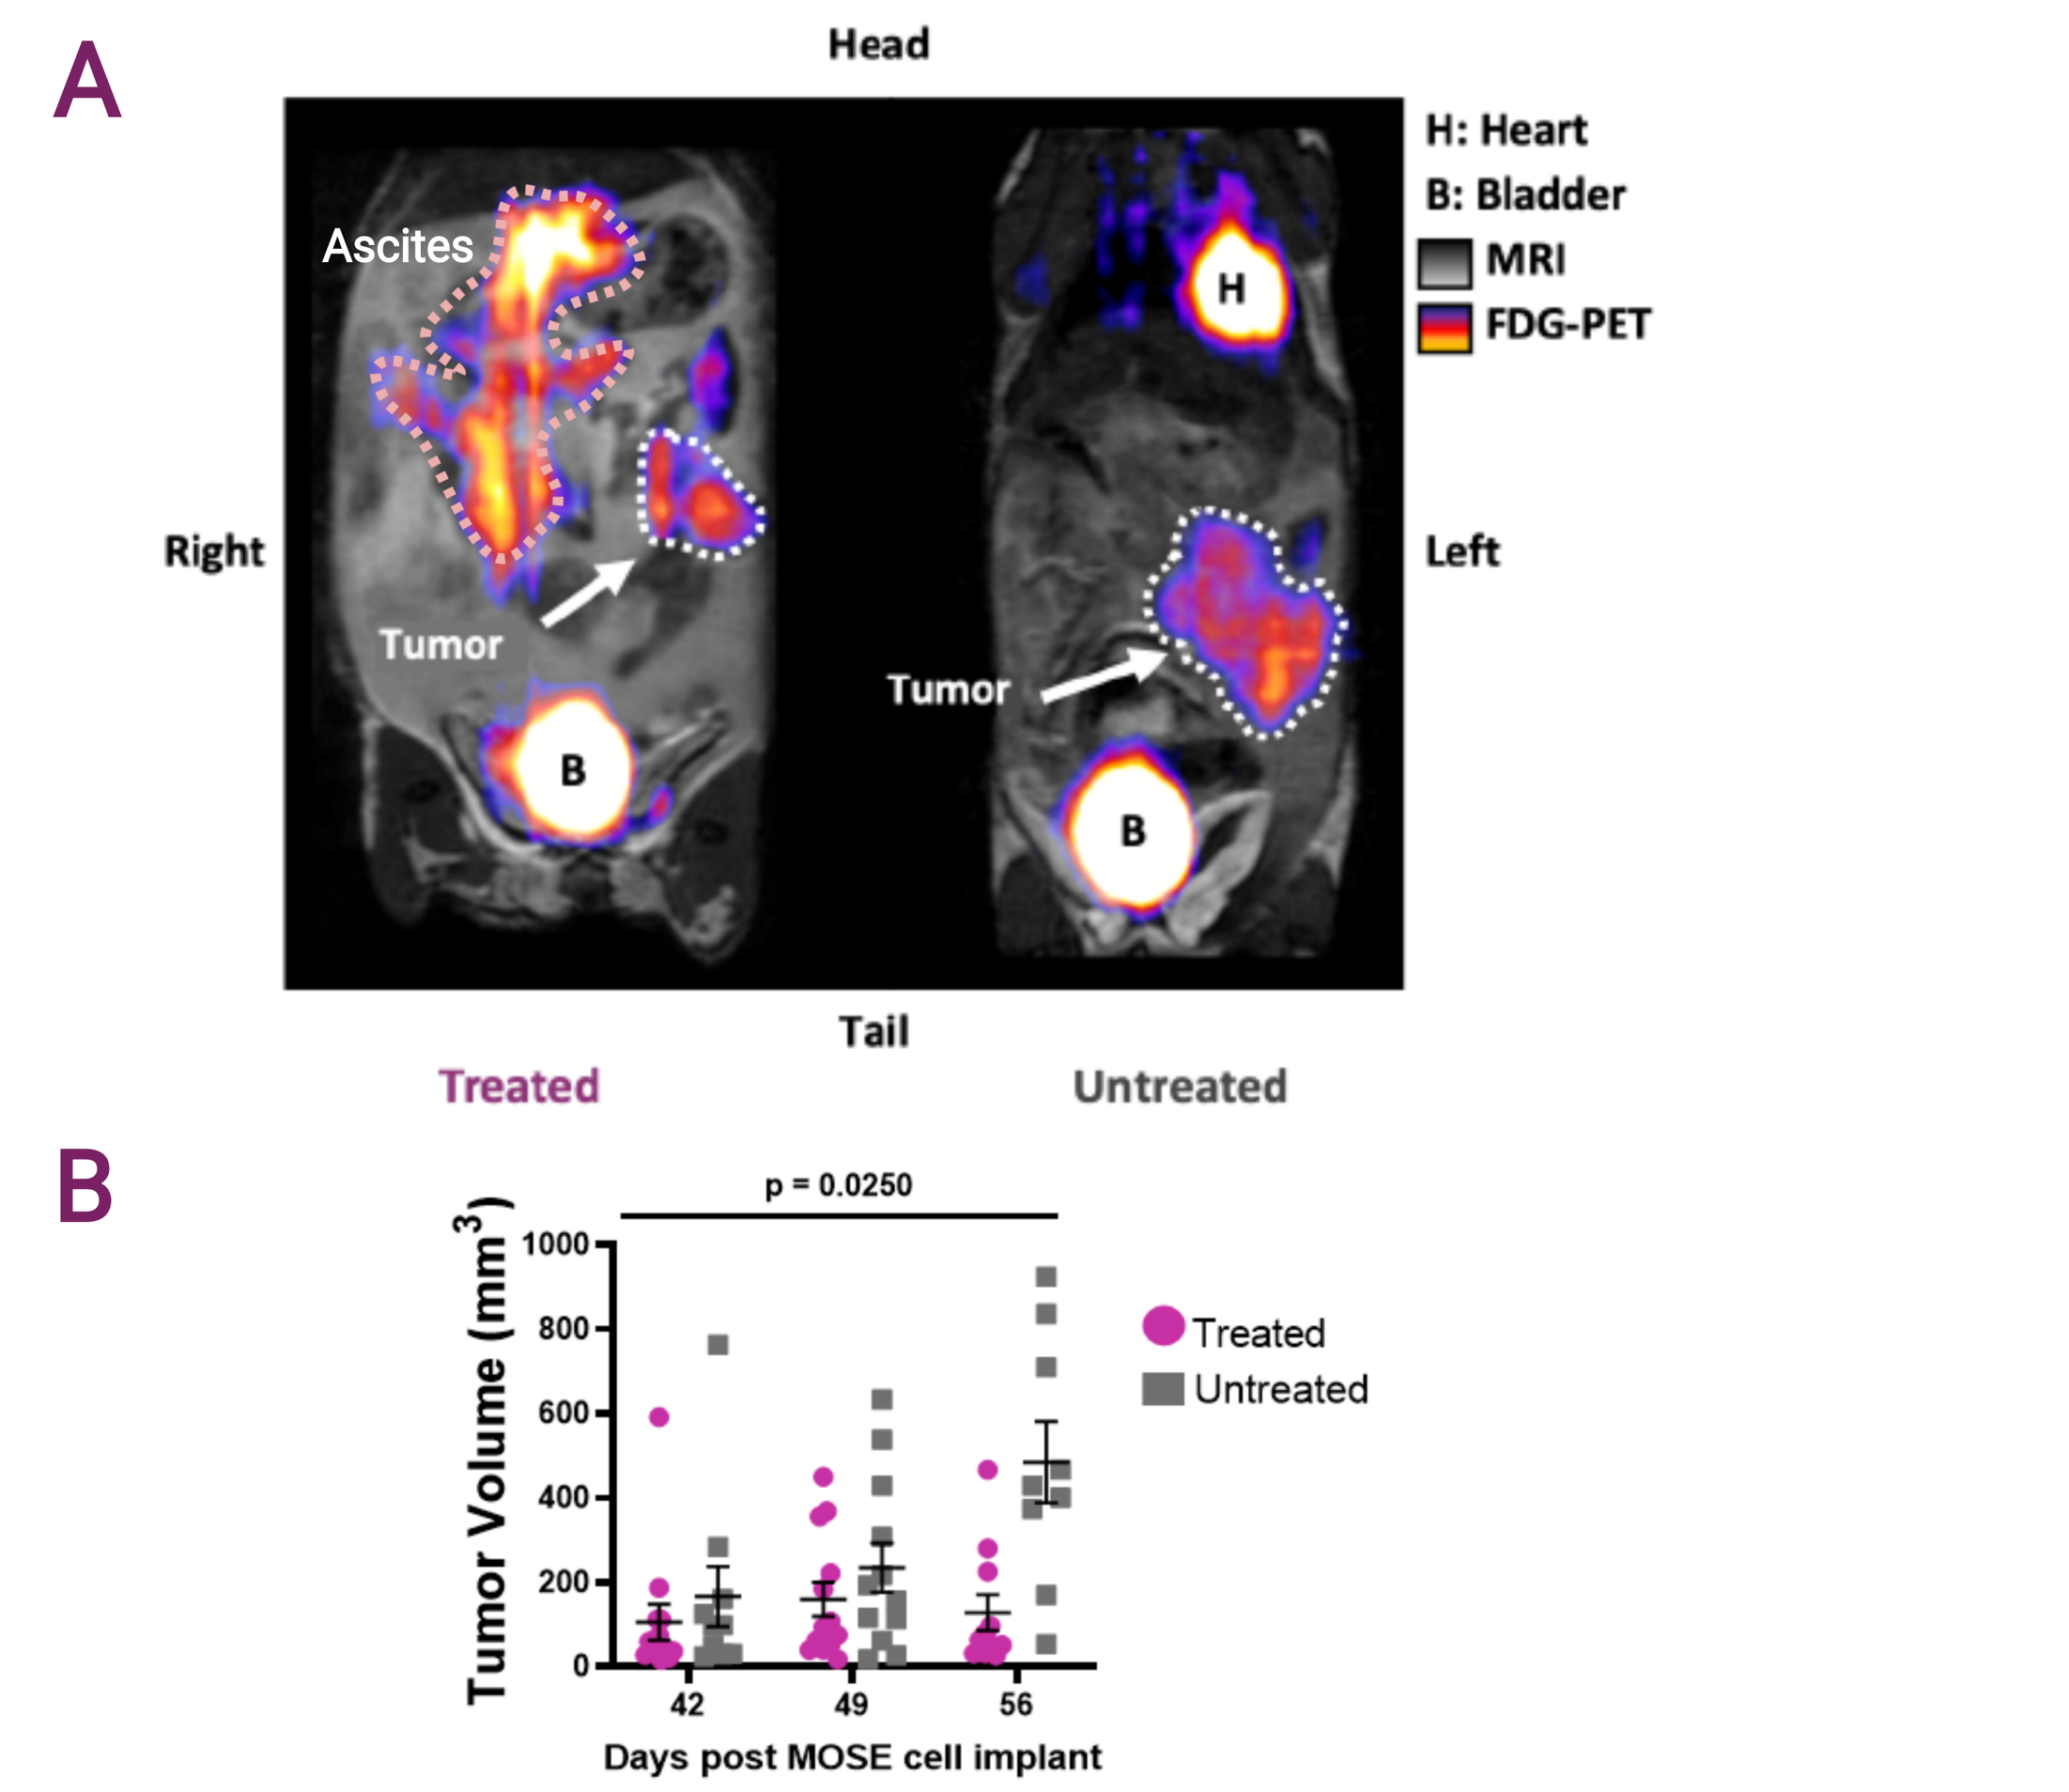
A
Head
H: Heart
B: Bladder
MRI
FDG-PET
Ascites
Right
Left
Tumor
Tail
Treated
Untreated
B
p = 0.0250
Tumor Volume (mm³)
Treated
Untreated
Days post MOSE cell implant

Figure 1: Treatment with DPX-Survivac/CPA and anti-PD-1 decreases tumor volumes. Tumor sizes were assesed by PET/MRI. **A)** Examples of untreated (right) and treated (left) mice on day 42. This treated mouse had a smaller tumor but also ascites throughout the abdominal cavity. **B)** On day 56, treated mice had smaller tumors whereas tumor volume increased from day 42 and 56 in untreated mice. The impact of treatment on mice survival could not be fully assessed due to the required termination of the mice on day 60 for the next studies.

Analysis of Ascites

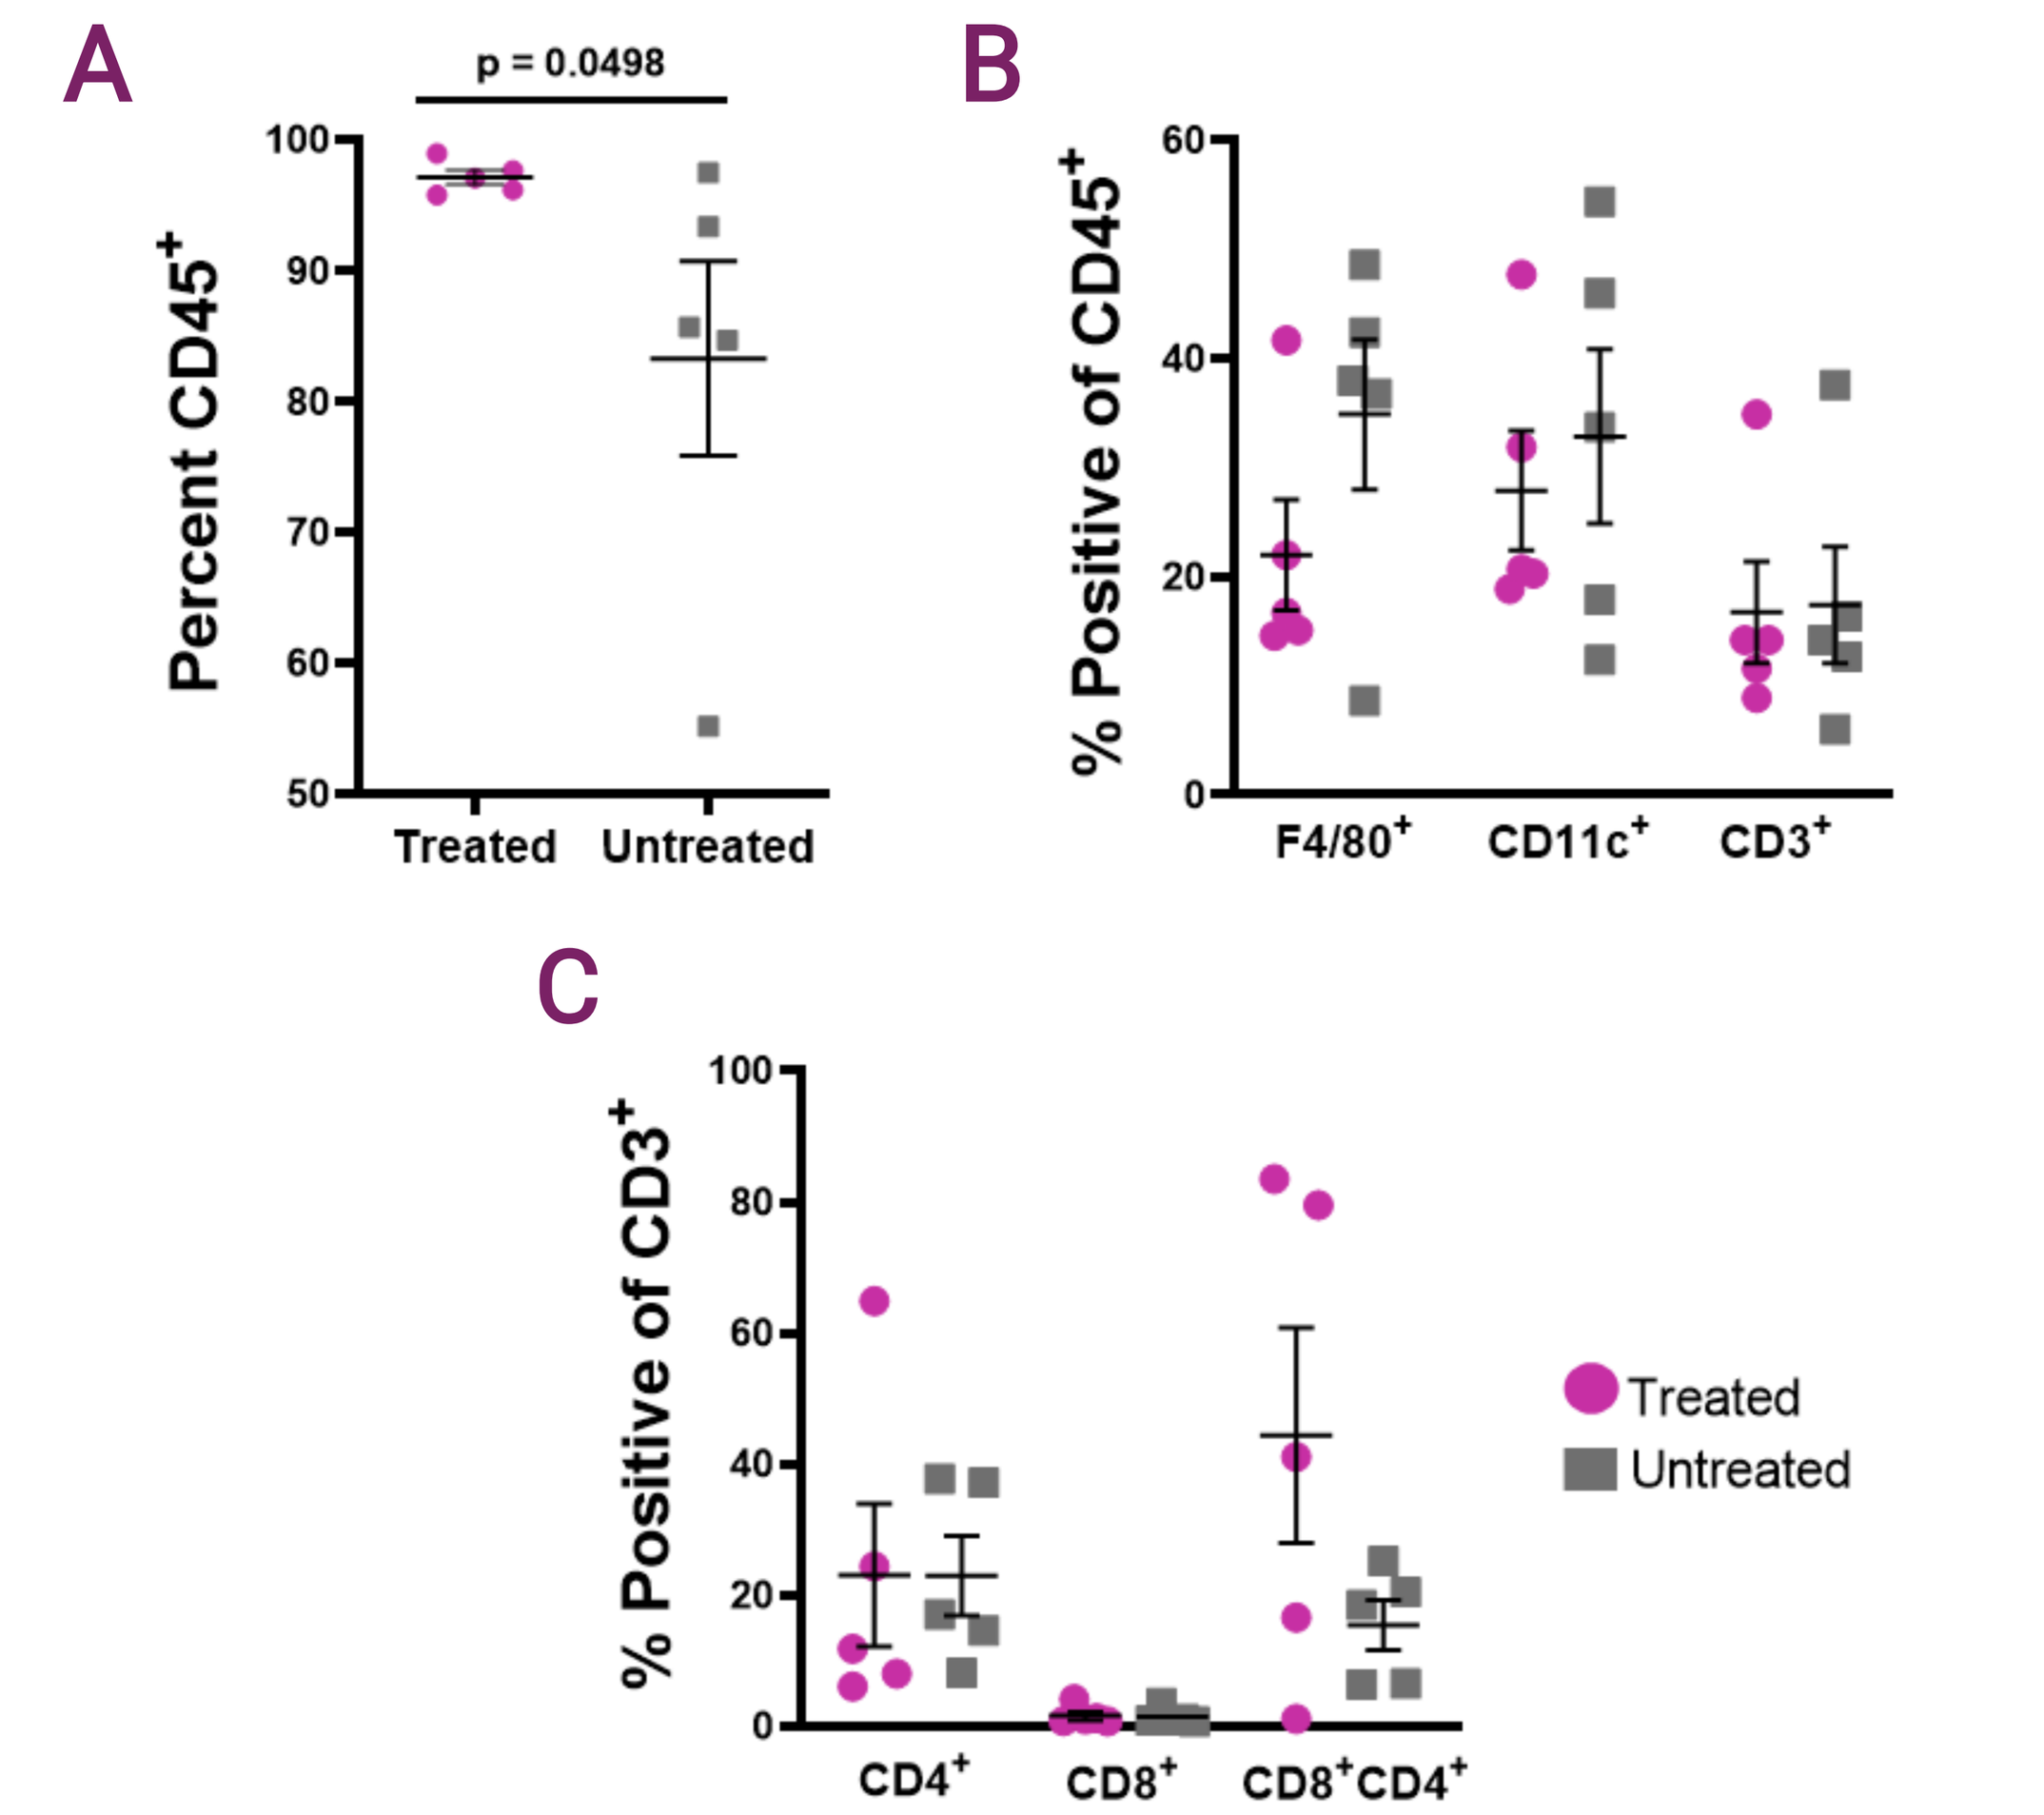
A
p = 0.0498
Percent CD45⁺
Treated
Untreated
B
% Positive of CD45⁺
F4/80⁺
CD11c⁺
CD3⁺
C
% Positive of CD3⁺
CD4⁺
CD8⁺
CD8⁺CD4⁺
Treated
Untreated

Figure 2: Treatment with DPX-Survivac/CPA and anti-PD-1 increases lymphocytes in ascites. 65% of treated and untreated mice had ascites. **(A)** Treated mice had significantly more $CD45^+$ cells than untreated mice. **(B)** Untreated mice appeared to have higher, percentages of $F4/80^+$ cells. No difference was observed for $CD11c^+$ or $CD3^+$ cells. **(C)** Treated mice appeared to have more $CD4^+CD8^+$ cells than untreated mice. No difference was observed for $CD4^+$ or $CD8^+$ cells.

Phenotyping Immune Cell Cultures

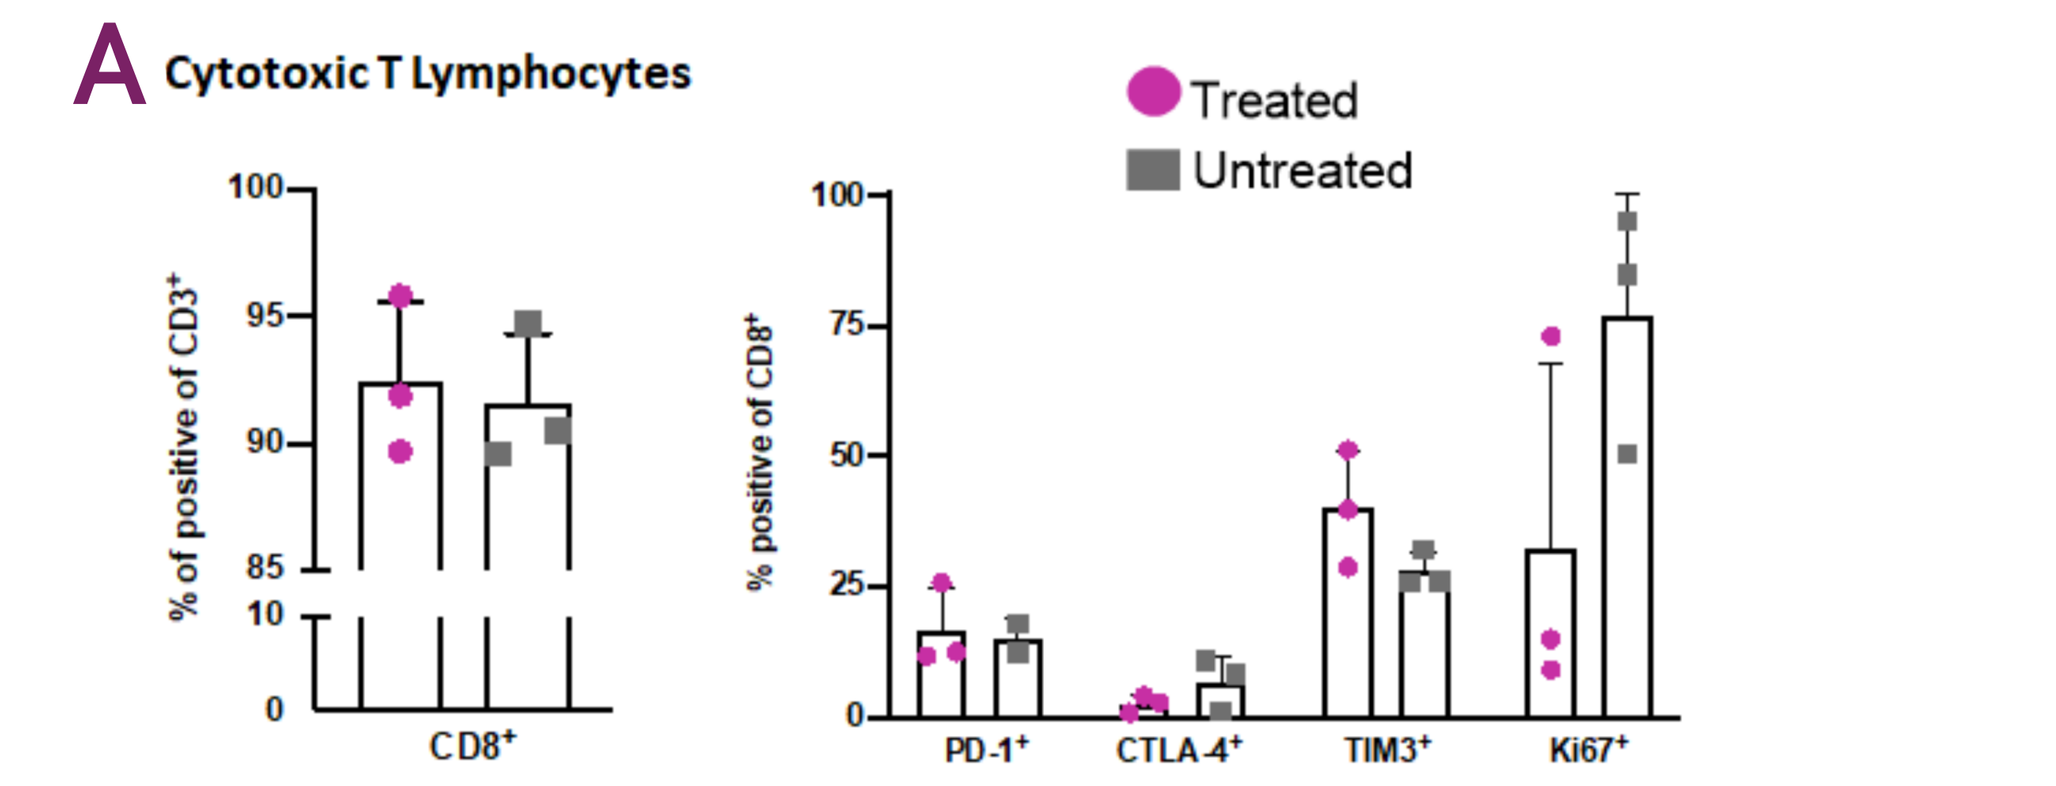
A Cytotoxic T Lymphocytes
Treated
Untreated
% of positive of CD3⁺
CD8⁺
% positive of CD8⁺
PD-1⁺
CTLA-4⁺
TIM3⁺
Ki67⁺

B Myeloid Cells

		Treated		Untreated	
		MHCII+ F4/80^high (Dendritic cells)	MHCII- F4/80^low (Macrophages)	MHCII+ F4/80^high (Dendritic cells)	MHCII- F4/80^low (Macrophages)
	Proportion of Total sample (%)	52.6	31.7	33.8	47.1
Proportion of MHCII + F4/80^high/low (%)	CD11c	96.8	22.8	88.7	41.7
	F4/80	100	82.8	100	62.9
	MHCII	100	5.36	79.1	12.4
	CD11b	99.9	99.9	100	99.6
	CD83	0.93	1.26	2.50	5.89
	CD8α	0.59	0.11	0.19	0.49

Figure 3: Treatment with DPX-Survivac/CPA and anti-PD-1 affects the phenotype of CTL and MC cultures. (A) CTL cultures from treated and untreated mice were very pure $CD3^+CD8^+$ cells with similar levels of PD-1. However, DPX/CPA and anti-PD-1 treatment increased expression of checkpoint inhibitor TIM3 and decreased expression of proliferation marker Ki67. **(B)** MC cultures from treated mice had an increased proportion of dendritic cells (DCs) compared to macrophages. Both cell types from treated mice had decreased expression of CD83, while MHCII expression decreased in macrophages but increased in DCs.

Cytotoxic T Lymphocyte and Myeloid Cell Recruitment to Tumors and Inguinal Lymph Nodes

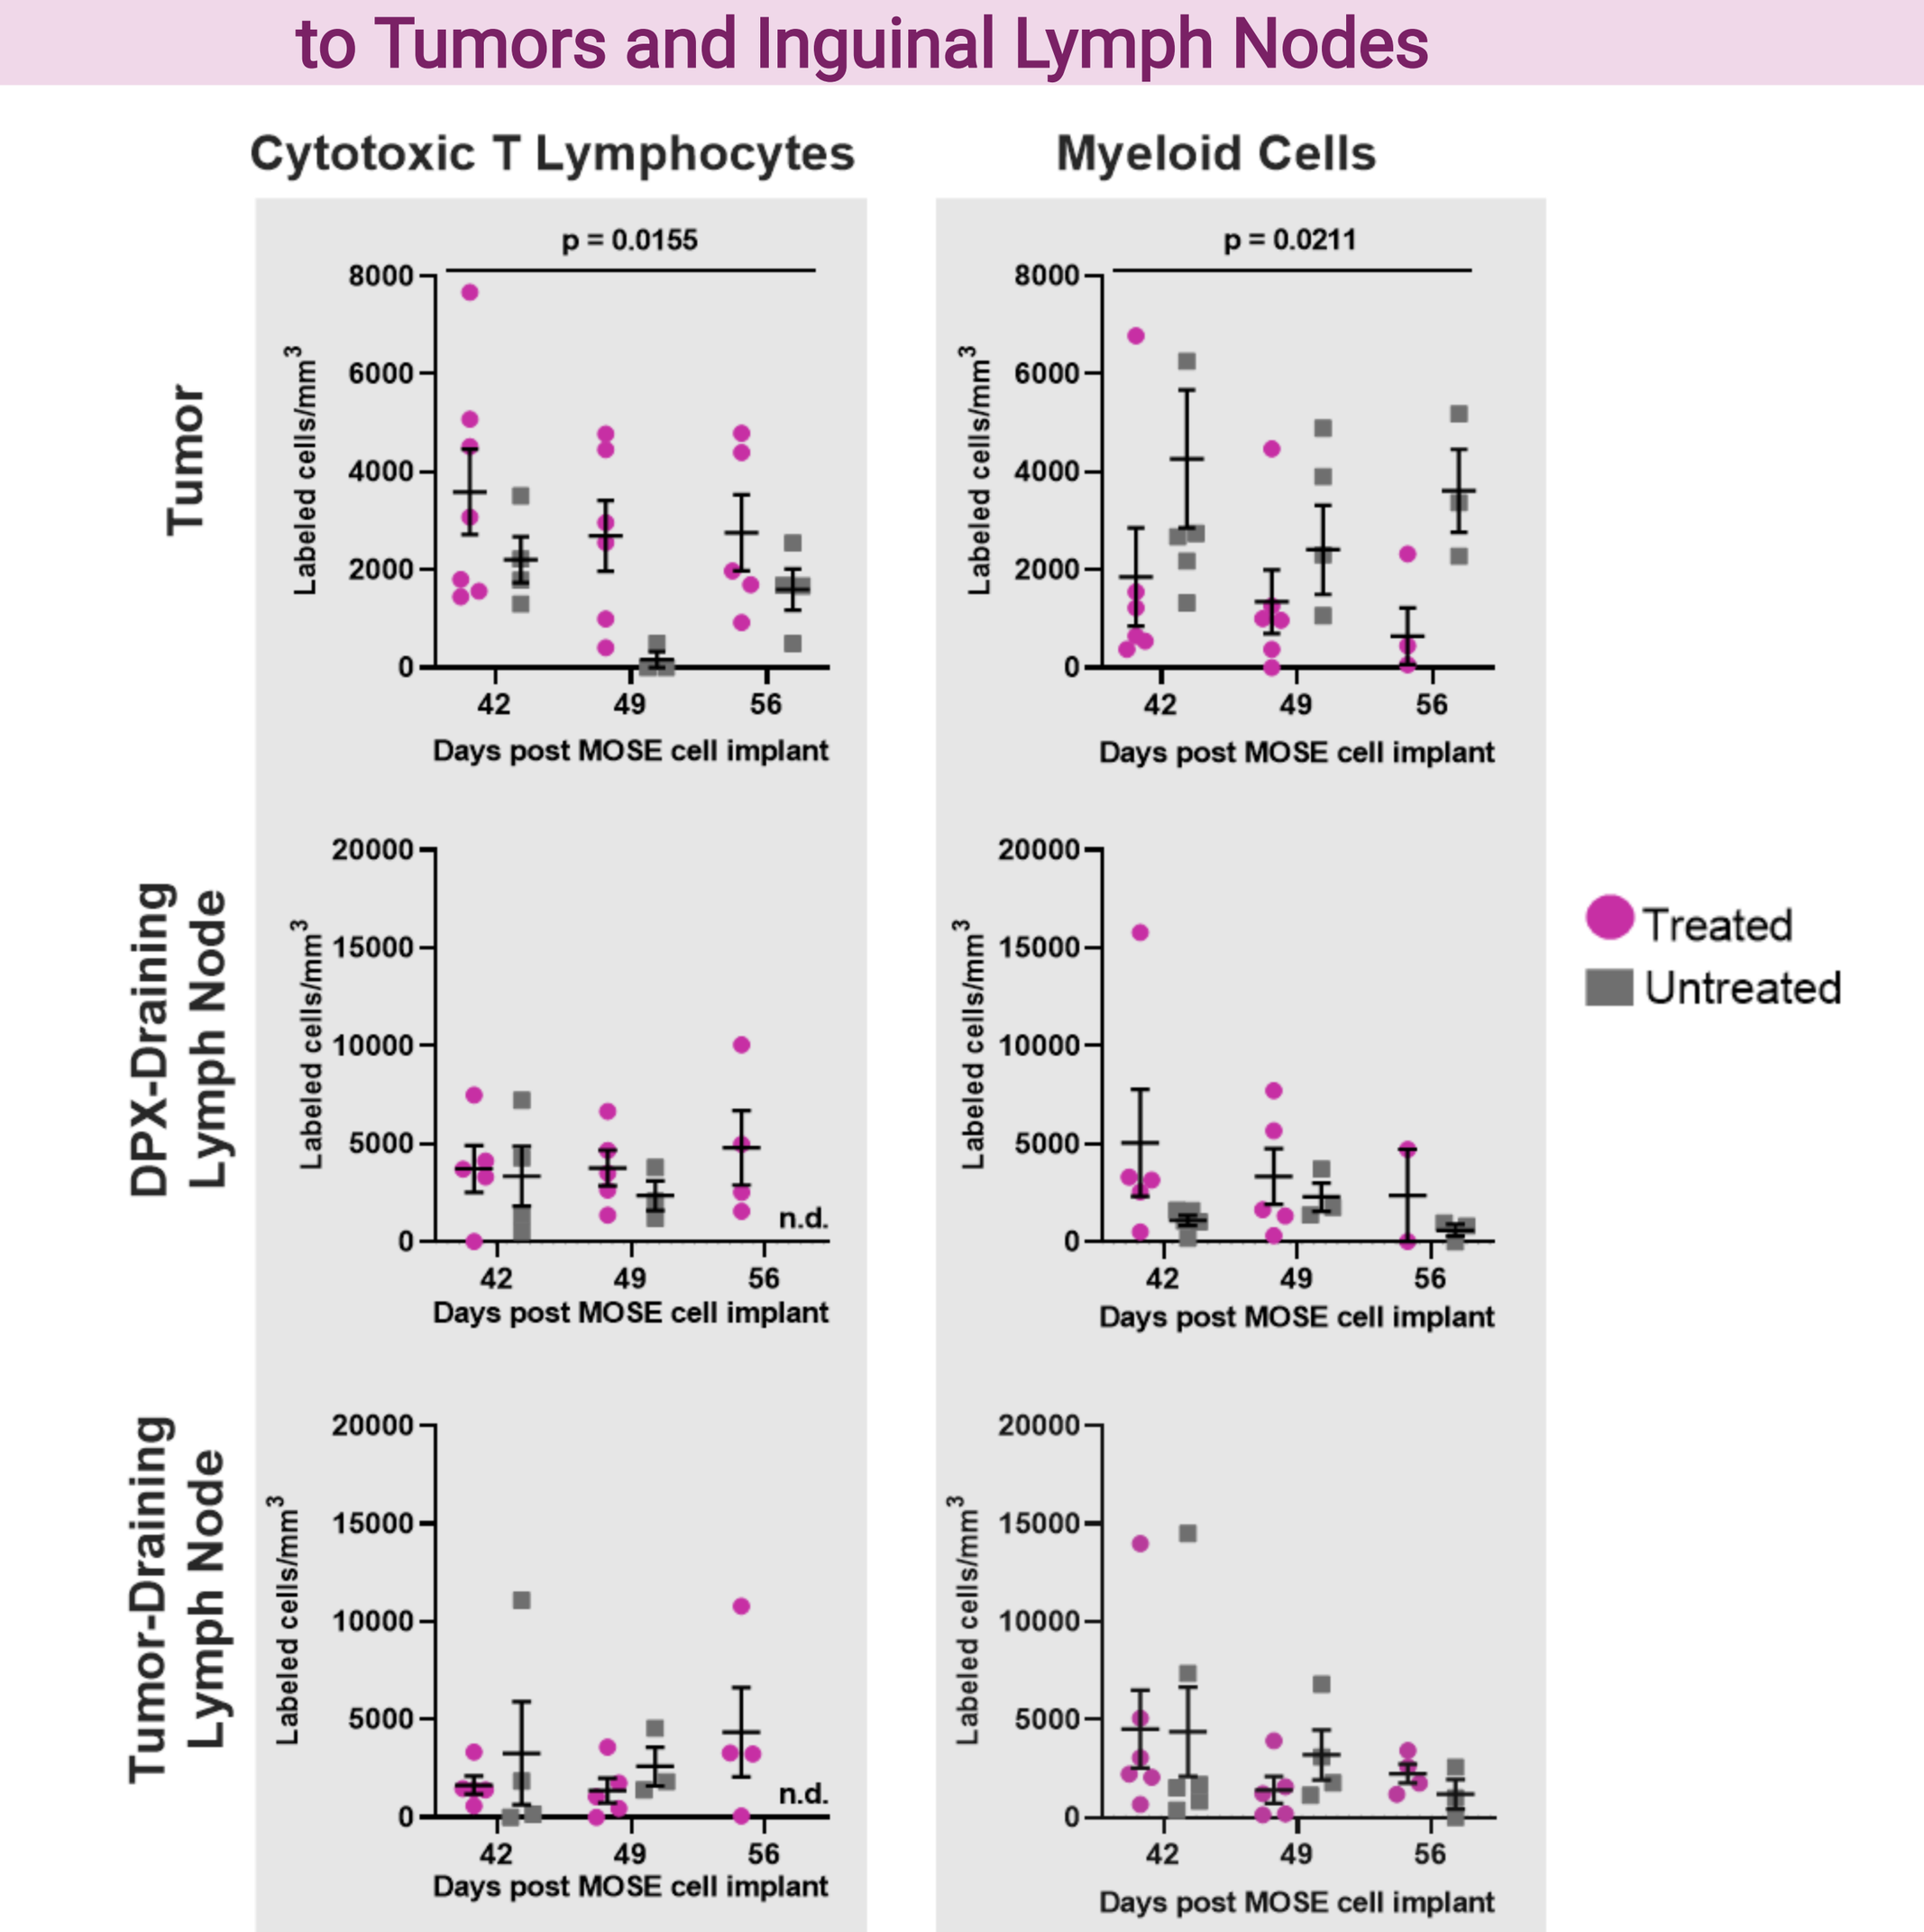
Cytotoxic T Lymphocytes
Myeloid Cells
Tumor
DPX-Draining Lymph Node
Tumor-Draining Lymph Node
p = 0.0155
p = 0.0211
Labeled cells/mm³
Days post MOSE cell implant
n.d.
Treated
Untreated

Figure 4: Treatment with DPX-Survivac/CPA and anti-PD-1 increases recruitment of CTLs into tumors. The number of CTLs & MCs recruited into the tumor and tumor-draining and DPX-draining lymph nodes was measured. In the tumor, recruitment of CTLs was significantly higher in treated mice, whereas untreated mice had higher MCs recruitment. In the DPX-draining lymph node, treated mice tended to have a higher number of cells recruited compared to untreated mice, albeit not statistically different. Treatment did not affect cell recruitment in the tumor-draining node.

Analysis of Tumor-Infiltrating Lymphocytes

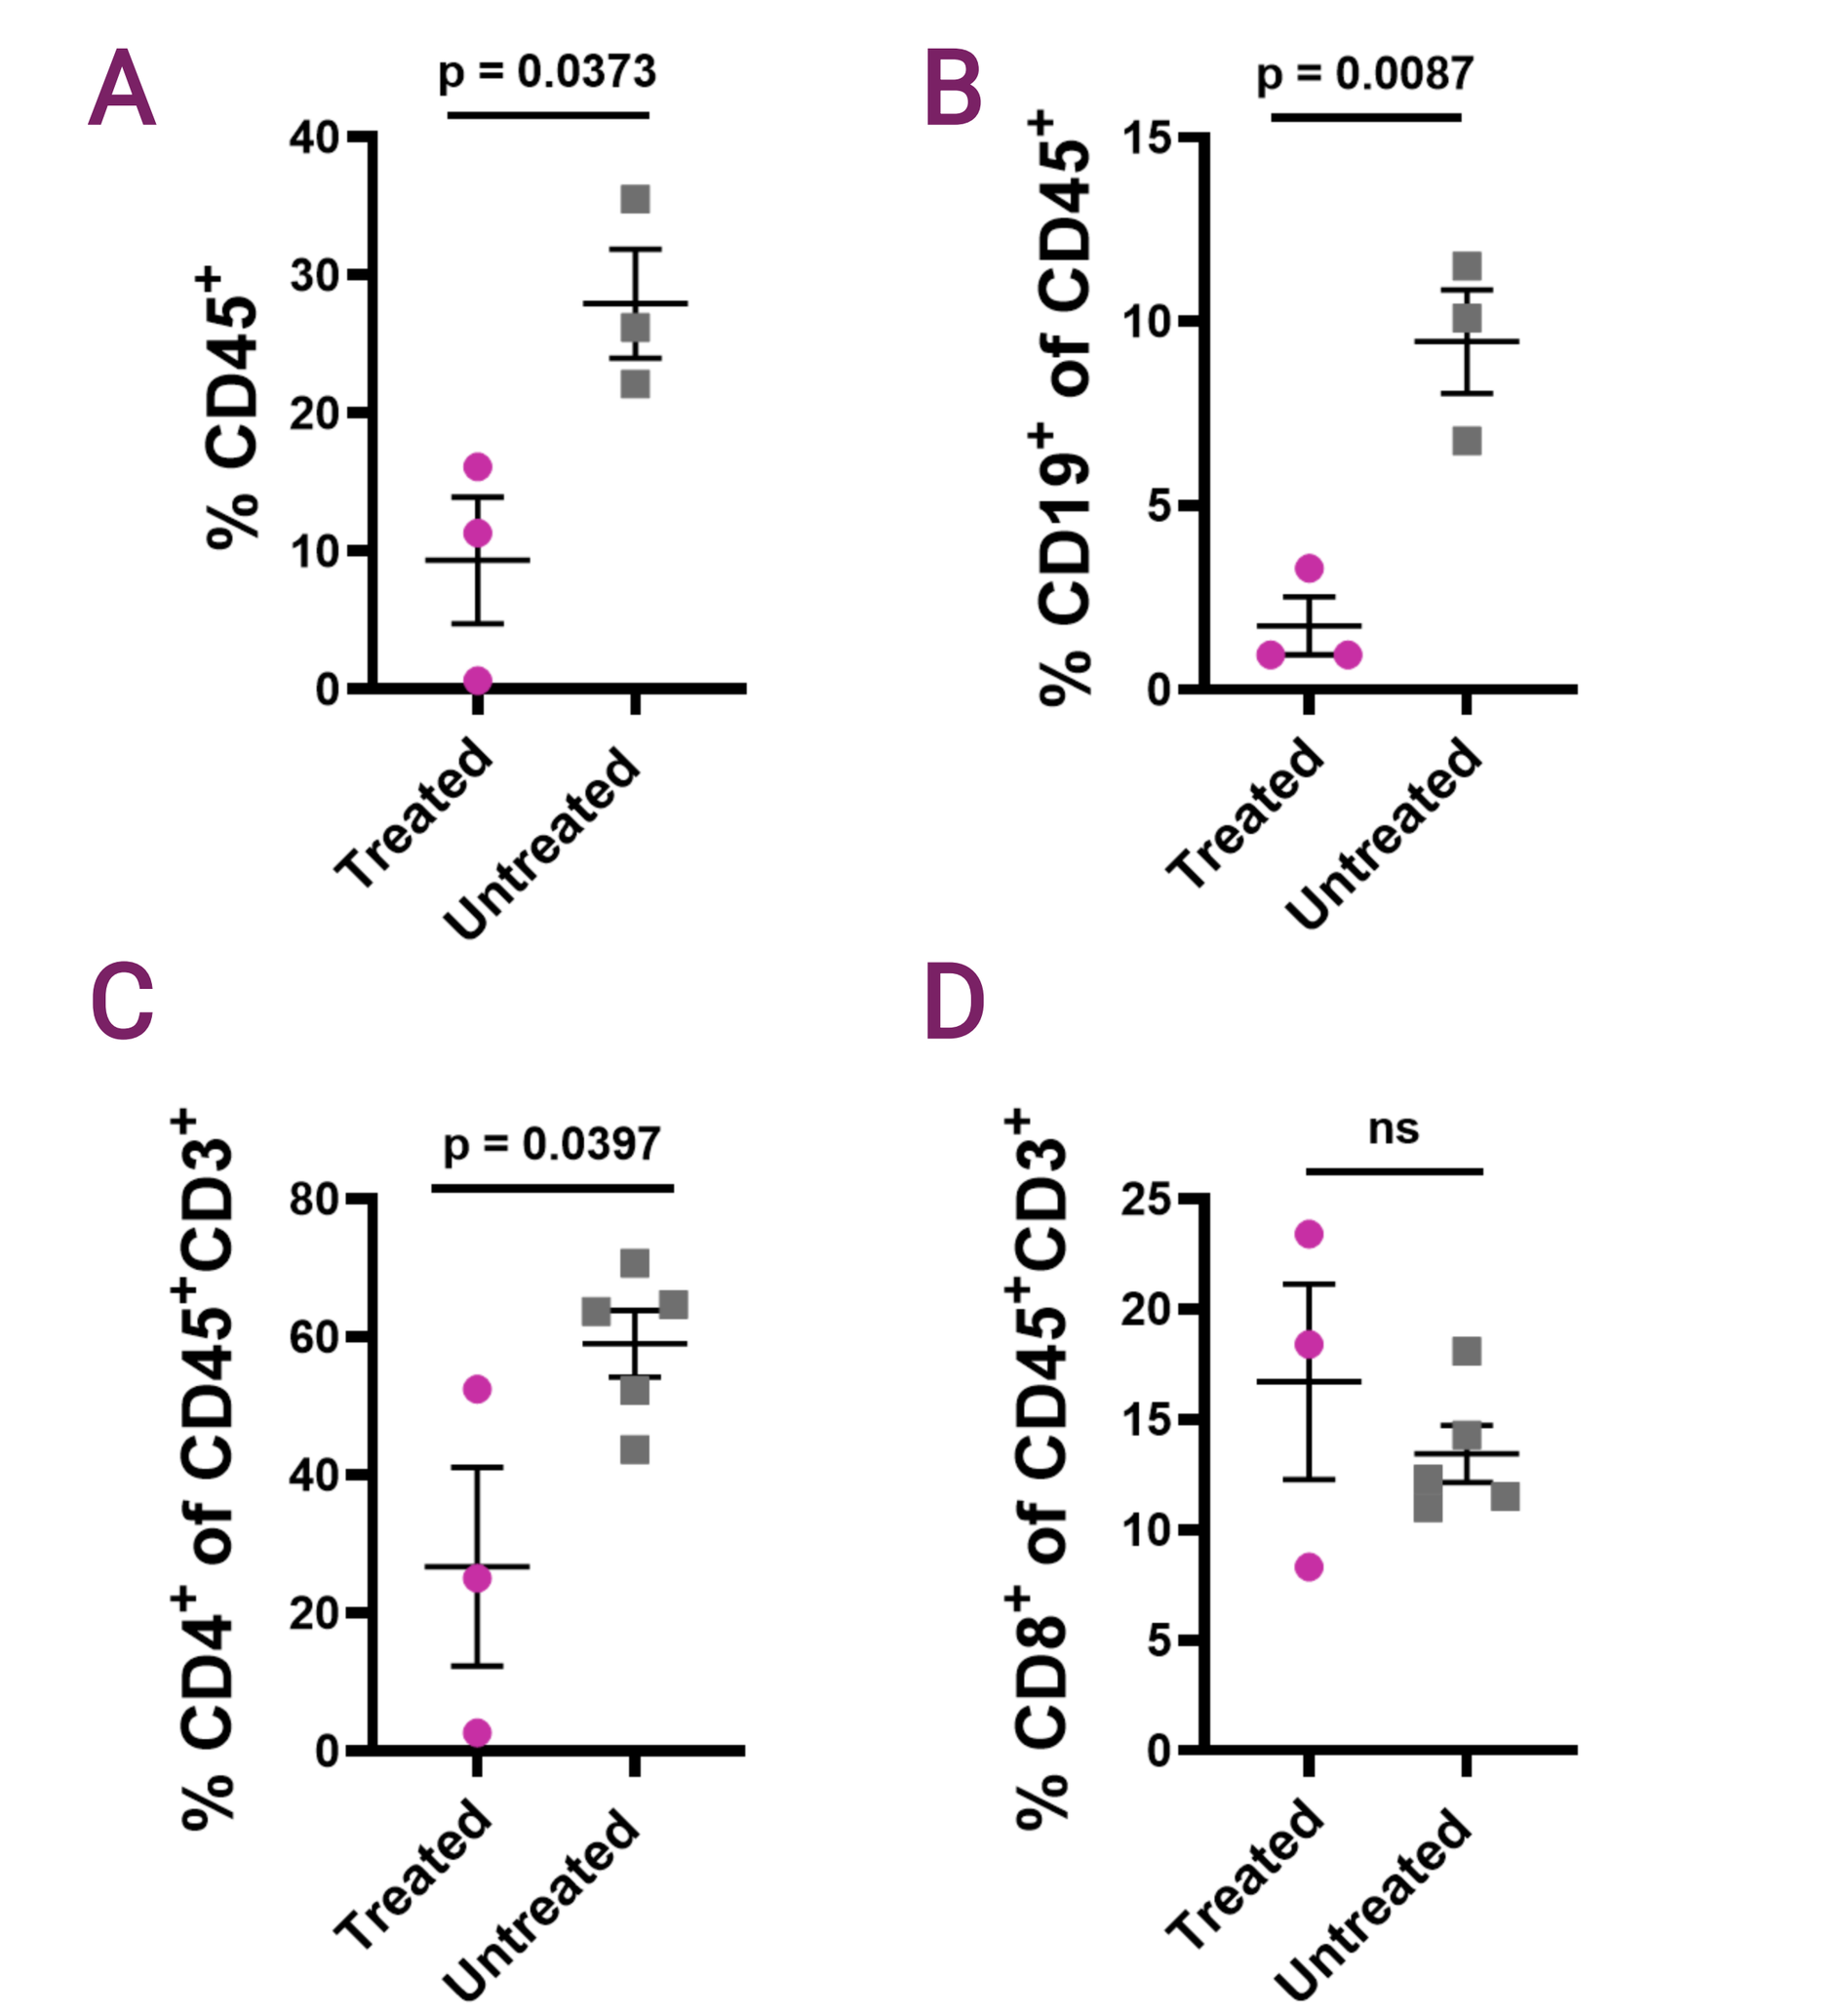
A
p = 0.0373
% CD45⁺
B
p = 0.0087
% CD19⁺ of CD45⁺
C
p = 0.0397
% CD4⁺ of CD45⁺CD3⁺
D
ns
% CD8⁺ of CD45⁺CD3⁺
Treated
Untreated

Figure 5: Treatment with DPX-Survivac/CPA and anti-PD-1 decreases number of B cells in tumors on day 56. Flow cytometry revealed that treated mice had significantly lower numbers of **(A)** $CD45^+$ lymphocytes, **(B)** $CD19^+$ B cells, and **(C)** $CD4^+$ T cells. However, there were no statistically significant changes in the number of **(D)** $CD8^+$ T cells. It should be noted that tumor volume was very heterogeneous within both treated and untreated groups.

Analysis of Suppressive Tumor Inflitrates

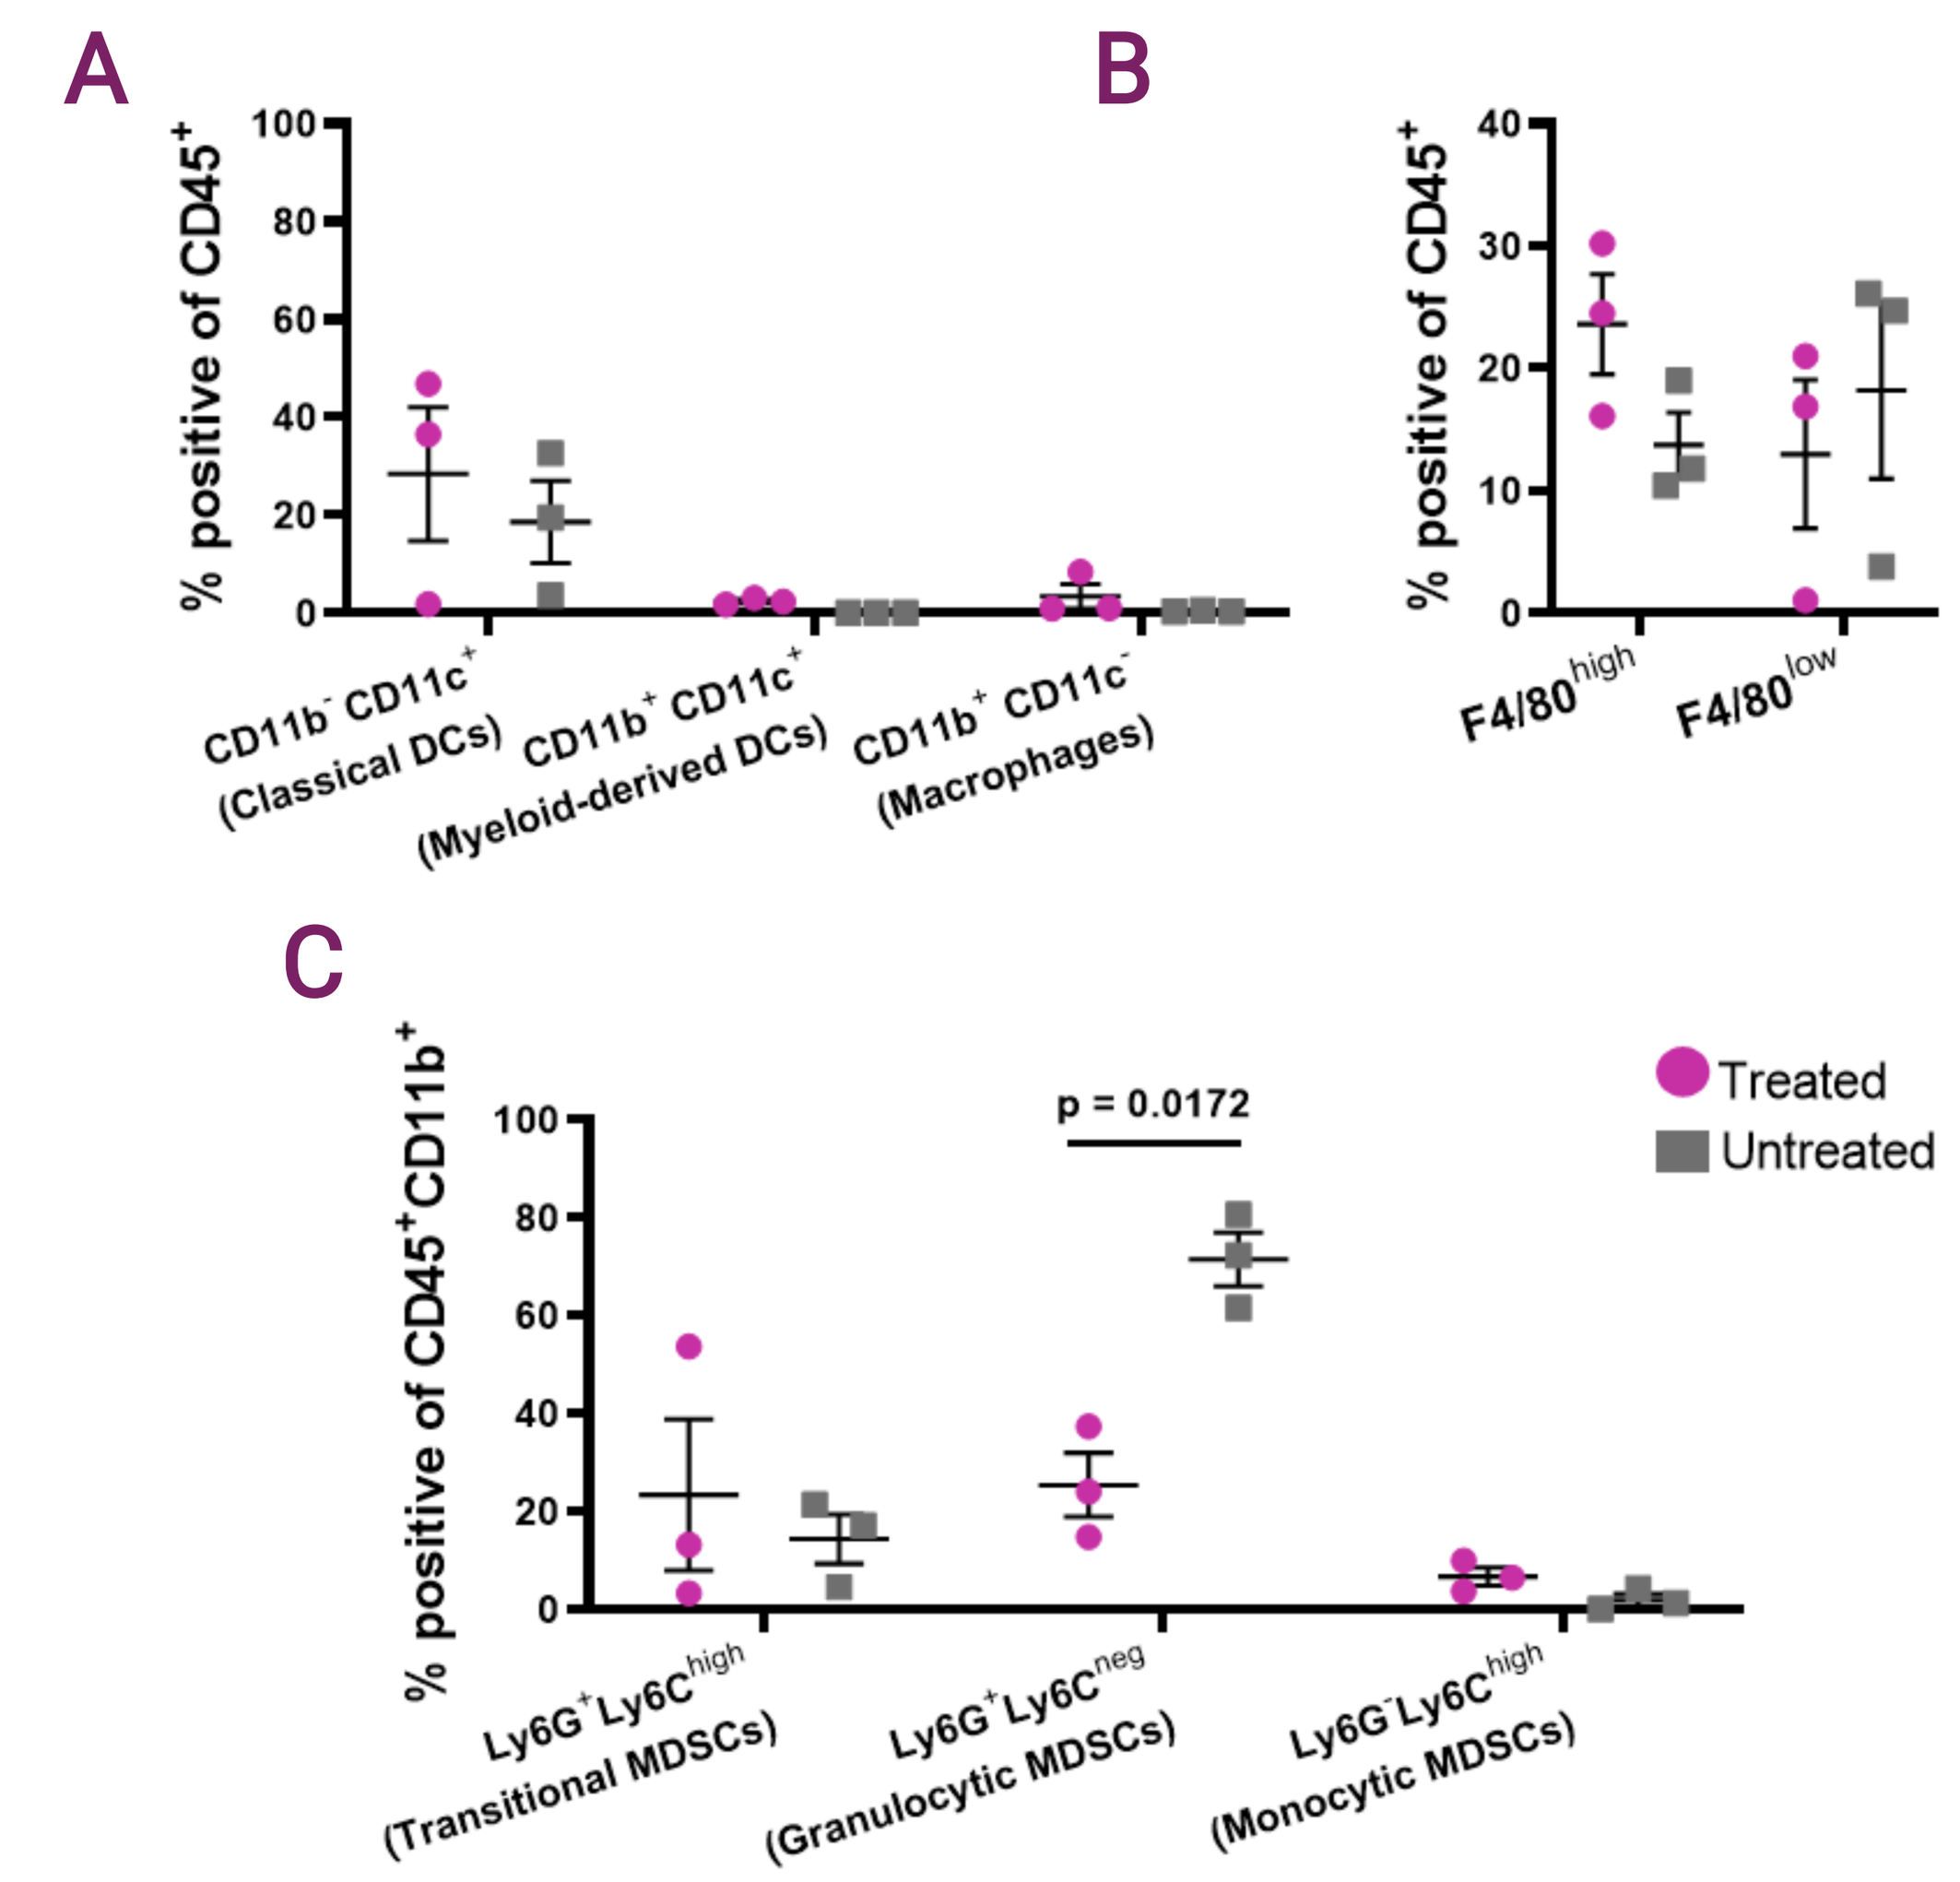
A
% positive of CD45⁺
CD11b⁻ CD11c⁺ (Classical DCs)
CD11b⁺ CD11c⁺ (Myeloid-derived DCs)
CD11b⁺ CD11c⁻ (Macrophages)
B
F4/80^high
F4/80^low
C
% positive of CD45⁺CD11b⁺
p = 0.0172
Ly6G⁺Ly6C^high (Transitional MDSCs)
Ly6G⁺Ly6C^neg (Granulocytic MDSCs)
Ly6G⁻Ly6C^high (Monocytic MDSCs)
Treated
Untreated

Figure 6: Treatment with DPX-Survivac/CPA and anti-PD-1 decreases granulocytic myeloid-derived suppressor cells (MDSCs) found in tumors on day 56. (A) No significant difference in the number of classical DCs ($CD11b^-CD11c^+$), myeloid-derived DCs ($CD11b^+CD11c^+$), or bone-marrow derived macrophages ($CD11b^+CD11c^-$) cells was observed, nor in **(B)** $F4/80^{high/low}$ cells ± treatment. **(C)** The number of $Ly6G^+Ly6C^{lo}$ cells (granulocytic MDSCs) was significantly decreased in treated mice, but no difference was found for other MDSC types.

CONCLUSIONS

In an orthotopic epithelial ovarian cancer mouse model, when compared to untreated, **DPX-Survivac/CPA/anti-PD-1 treatment resulted in:**

* **Primary tumor control**

* **Higher migration of $CD8^+$ T cells into tumors** (as assessed via MRI)

* **Trend for an increased recruitment of $CD8^+$ T cells and myeloid cells to the DPX-draining lymph node** (as assessed via MRI)

* **Increased numbers of $CD45^+$ lymphocytes in the ascitic fluid**

* **Reduced numbers of $CD19^+$ B cells and granulocytic MDSCs ($Ly6G^+Ly6C^{lo}$) in tumors** on day 56 (as assessed via flow cytometry)

Further information: The authors would like to acknowledge funding support from MITACS and the Beatrice Hunter Cancer Research Institute (BHCRI)

Disclosure: At time of study, MT, AM, KBo, RB, GW, AMJ and MS were employees of IMV Inc., and KBr had contracts with IMV for a separate project.

Acknowledgements: The authors would like to thank Dr. Rajkannan Rajagopalan for assistance with vaccine preparation and Dr. Brennan Dirk with flow cytometry analysis.