RX-5902 Exhibits Direct & Immunomodulatory Anti-tumor Activities In Melanoma PDX Models

Andrew Eisen
Rexahn Pharmaceuticals, Inc.

#4127

Background & Methods

Background: Nuclear β-catenin promotes deleterious processes in tumors and their microenvironment. Diminishing nuclear β-catenin attenuates these oncogenic processes. RX-5902 is an oral, small molecule inhibitor of β-catenin nuclear translocation mediated by phosphorylated p68 (DDX5). RX-5902 exhibits potent anti-cancer activity in many cancer cell lines, xenograft models in immunodeficient mice and immune competent mice. This study examines the relative contribution and context of the direct and immunomodulatory effects on the anti-tumor activities of RX-5902.

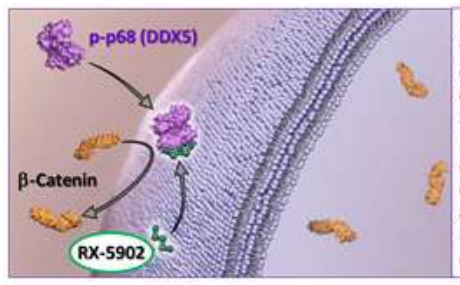
p-p68 (DDX5)
β-Catenin
RX-5902

RX-5902 binds to p-p68 and disrupts the formation of a protein complex with β-catenin. This inhibits the transportation of β-catenin across the nuclear membrane and leads to a reduction in β-catenin in the nucleus. With markedly reduced nuclear β-catenin, oncogenes are turned off in cancer cells, inhibiting tumor growth. Immune cells are then activated against the tumor cells - providing a two-pronged attack on the cancer cells.

Methods: Four patient-derived melanoma xenograft models were implanted into either immunodeficient nude mice or NOG mice "humanized" with HLA unmatched CD34+ stem cells with average humanization of 36%. The PDX models were derived from 2 tumors that were clinically unresponsive to an anti-PD-1 agent and 2 tumors obtained prior to any immune treatment. In each category one tumor had BRAF WT and one a V600E mutation. Mice were randomized to receive either vehicle or RX-5902 at 5, 15 or 50 mg/kg po QDx5/Off x2 when implanted tumors reached 100-120 mm³ and began treatment when tumors reached 200mm³. Treatment lasted for 28 days and animals were observed for an additional 14 off therapy.

Study Design

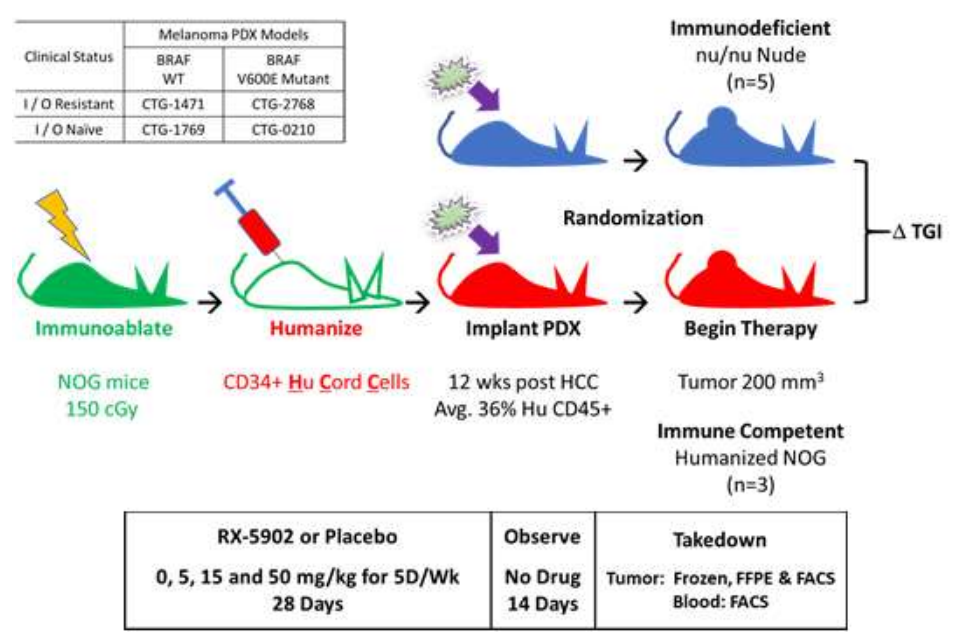
Melanoma PDX Models
Clinical Status
BRAF WT
BRAF V600E Mutant
I / O Resistant
CTG-1471
CTG-2768
I / O Naive
CTG-1769
CTG-0210
Immunoablate
NOG mice
150 cGy
Humanize
CD34+ Hu Cord Cells
Implant PDX
12 wks post HCC
Avg. 36% Hu CD45+
Randomization
Immunodeficient
nu/nu Nude
(n=5)
Begin Therapy
Tumor 200 mm³
Immune Competent
Humanized NOG
(n=3)
Δ TGI
RX-5902 or Placebo
0, 5, 15 and 50 mg/kg for 5D/Wk
28 Days
Observe
No Drug
14 Days
Takedown
Tumor: Frozen, FFPE & FACS
Blood: FACS

TGI vs RX-5902 Dose

Tumor Growth Inhibition vs Dose At Days 28, 35 & 42
(Relative to Placebo)

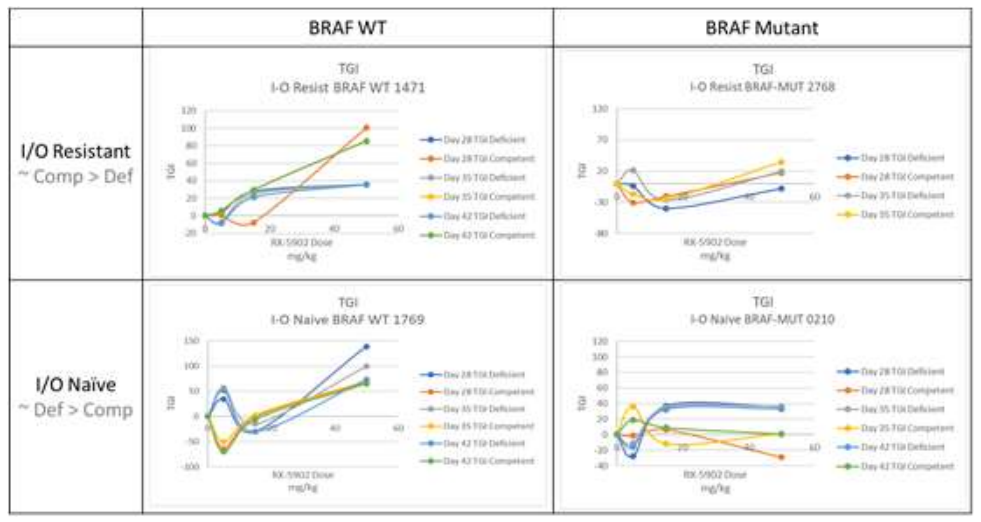
BRAF WT
BRAF Mutant
I/O Resistant
~ Comp > Def
I/O Naive
~ Def > Comp
TGI
I-O Resist BRAF WT 1471
TGI
I-O Resist BRAF-MUT 2768
TGI
I-O Naive BRAF WT 1769
TGI
I-O Naive BRAF-MUT 0210
RX-5902 Dose
mg/kg

Primary Outcome: Day 28 ΔTGI

Average Melanoma PDX Tumor Growth Inhibition (%)

Clinical Category		I/O Resistant		I/O Naïve	
BRAF Status		WT	V600E	WT	V600E
Day 28 TGI (On Drug)	Immunocompetent	100	17	67	-29
	Immunodeficient	35	-8	138	36
Day 35 TGI (Off Drug)	Immunocompetent	85	34	70	1
	Immunodeficient	30	20	99	36
Champions Oncology Models		CTG-1471	CTG-2768	CTG-1769	CTG-0210

A Secondary Outcome At Day 42

Blood Phenotyping vs Dose at Day 42

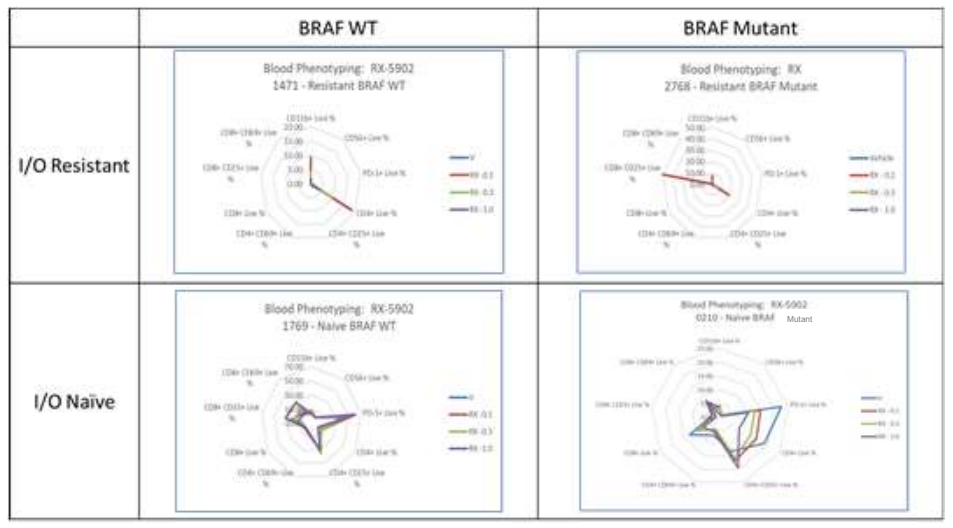
BRAF WT
BRAF Mutant
I/O Resistant
I/O Naïve
Blood Phenotyping: RX-5902
1471 - Resistant BRAF WT
Blood Phenotyping: RX
2768 - Resistant BRAF Mutant
Blood Phenotyping: RX-5902
1769 - Naive BRAF WT
Blood Phenotyping: RX-5902
0210 - Naive BRAF Mutant

Conclusions

- RX-5902 has been shown to augment anti-PD-1 immunotherapy in multiple pre-clinical cancer models (syngeneic and humanized), including triple-negative breast cancer, when used in combination therapy.
- This study provides a comparison of tumor growth inhibition (ΔTGI) of RX-5902 monotherapy in 4 patient-derived melanoma xenograft (PDX) models in both immune deficient and immune competent mice to identify RX-5902-induced anti-tumor immune activity during and following therapy.
- RX-5902 provided greater tumor growth inhibition (ΔTGI) in the two melanoma PDX models derived from subjects who were resistant to I/O therapy compared to the two models from I/O naïve subjects.
- TGI is greater in BRAF WT vs BRAF V600E mutant tumors.
- RX-5902 immune-mediated anti-tumoral activity may benefit the majority of melanoma subjects with limited response to BRAF-directed and/or I/O therapy.

Acknowledgements

This study was designed and supported by Rexahn Pharmaceuticals. Work was conducted at Champions Oncology under the direction of Dr. D. Shah and Mr. Lonnie Myer.

rexahn PHARMACEUTICALS