Phenotyping Pancreatic Cancer CTCs As Biomarkers For RX-3117 Clinical Trials

D.J. Kim[1], Jie Yang[2], Weiguo Wu[2], Darren W. Davis[2] & Andrew Eisen[1]
[1]Rexahn Pharmaceuticals, Inc. and [2]Precision for Medicine, Houston, TX

#428

Background & Methods

Background:
RX-3117 is an oral, small molecule nucleoside analogue pro-drug being used in combination with nab-paclitaxel in a Ph2 clinical trial for pancreatic cancer. RX-3117 can be transported intracellularly by SLC29A1 (hENT1) and is converted to an active agent by the cancer-enriched enzyme, uridine-cytidine kinase 2 (UCK2) for incorporation into RNA and DNA. This leads to cancer cell apoptosis and epigenetic effects. We developed an approach to enumerate and phenotype circulating tumor cells (CTCs) from pancreatic cancer subjects by quantitative immunofluorescence (QIF) to assess whether CTC numbers, phenotypic features and/or early apoptotic responses to therapy predict or presage clinical response.

Methods:
QIF staining parameters were developed with cancer cell lines sensitive and resistant to RX-3117 utilizing monoclonal antibodies to hENT1 and UCK2. The staining methods were then applied to CTCs isolated from 10 ml anticoagulated blood by dielectrophoretic (DEP) properties that differentiate cancer cells and normal PBMCs using the ApoStream device (ApoCell, TX). In addition to the drug-related markers, CTCs were stained for a panel of either Epithelial or Mesenchymal markers, CD45 and nuclei (DAPI). Phenotyped CTCs were binned into 6 categories defined by whether they were EPI+/-, EMT+/- or CD45+/-. The percentage of cells in each category also positive for hENT1 or UCK2 was determined as was the mean fluorescence intensity of the marker+ sub-population. In addition, assessment of hENT1 and UCK2 expression was performed by qRT-PCR utilizing mRNA isolated from isolated CTCs, the buffy coat from spun blood and in plasma.

CTC Isolation & Characterization

Methods Development With Cancer Cell Lines

Cell Lines	Antibodies	Total Mean Fluorescence UCK2 Mab Titrations (ug/ml)		
		6.5	13	26
Mixture HL60 (S) BT549 (R)	UCK2	21.80	52.03	156.98
	Control	21.42	24.	59.09
	Ratio (S/N)	1.02	2.12	2.66

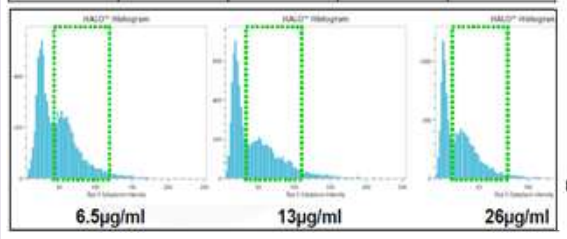
6.5µg/ml
13µg/ml
26µg/ml

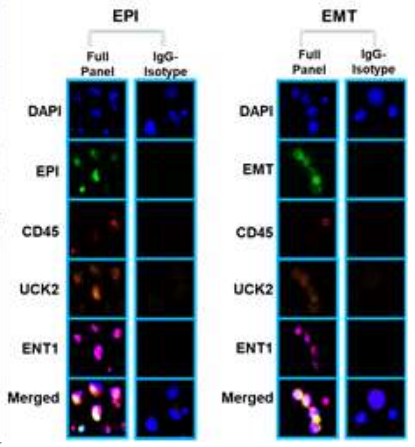
EPI
Full Panel
IgG-Isotype
DAPI
EPI
CD45
UCK2
ENT1
Merged
EMT
Full Panel
IgG-Isotype
DAPI
EMT
CD45
UCK2
ENT1
Merged

Cancer cells and CTCs are phenotyped for epithelial (EPI+) or mesenchymal (EMT+) markers. EPI markers include CK (AE1/AE3), CK (C11), EpCAM, E-Cadherin & β-catenin. EMT markers include Vimentin, N-Cadherin, TWIST, CEA, CA19-9 & CA125. HL60 is sensitive (S) and BT549 is resistant (R) to RX-3117.

CTC Phenotypes In PDAC Patients

CTCs Are Isolated, Enumerated (#/ml) & Phenotyped

Patient ID	EPI+/CD45-	EPI+/CD45+	EPI-/CD45-	EMT+/CD45-	EMT+/CD45+	EMT-/CD45-
120651224	321.50	368.88	344.75	244.75	353.63	175.88
120785056	4,951.38	5,580.25	124.50	4,739.50	3,821.88	687.88
121005321	598.88	1,015.13	333.25	633.88	634.50	391.50
121070087	9.88	41.63	14.75	34.38	26.75	6.13
121047830	121.38	155.63	21.50	125.38	161.25	12.75

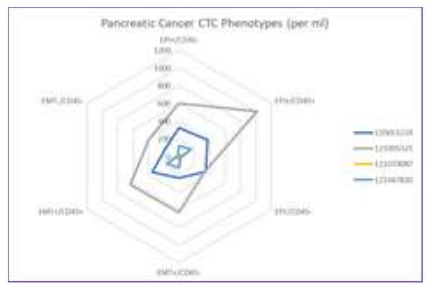
Pancreatic Cancer CTC Phenotypes (per ml)

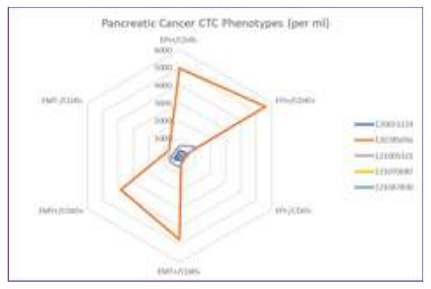
Pancreatic Cancer CTC Phenotypes (per ml)

Conclusions

- CTCs can be readily isolated, enumerated and characterized from PDAC patients by the ApoStream technology.
- In a pilot study of 5 PDAC subjects, CTC numbers ranged from 100 to 20, 000 / ml independent of phenotyping.
- CTCs can be phenotyped by expression of epithelial or mesenchymal markers and presence or absence of CD45.
- Assessment of biomarkers relevant to the RX-3117 MOA - hENT1 (SLC29A) and UCK2 - can be quantified by immunofluorescence.
 - The percentages of UCK2+ cells were higher in EPI+/EMT+ subsets than EPI-/EMT- subsets.
 - The percentages of hENT1+ cells were generally low in all of the subsets.
- This approach is now being used to monitor the response of PDAC patients in a Phase 2 trial of RX-3117 in combination with nab-paclitaxel (NCT03189914).

Study Design

Methods Development With Cancer Cell Lines

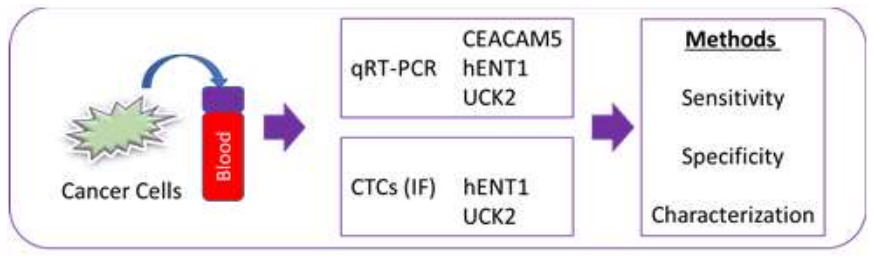
Cancer Cells
Blood
qRT-PCR
CEACAM5
hENT1
UCK2
CTCs (IF)
hENT1
UCK2
Methods
Sensitivity
Specificity
Characterization

Pilot Study In Independent PDAC Subjects

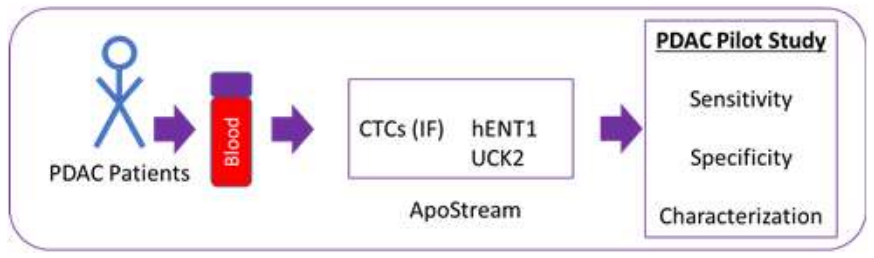
PDAC Patients
Blood
CTCs (IF)
hENT1
UCK2
ApoStream
PDAC Pilot Study
Sensitivity
Specificity
Characterization

PDAC Patient Demographics

PDAC Pilot CTC Study Participants

Patient ID	Current Age	Gender	Ethnicity	Primary Diagnosis	Treatment Status	Current Treatment	Past Treatment (where available)
120651224	68	Male	Caucasian	Stage IV Pancreatic Cancer	Active	FOLFIRI/Octreotide IM	Onivyde Leucovorin Fluorouracil Gemcitabine/Abraxane
120785056	67	Male	Caucasian	Stage IV Pancreatic Cancer	Active	Gemcitabine/Abraxane	FOLFIRINOX Irinotecan Oxaliplatin Leucovorin or Levoleucovorin FOLFIRI
121005321	50	Male	Caucasian	Stage IV Pancreatic Cancer	Active	FOLFIRINOX	Unknown
121070087	57	Male	Caucasian	Stage IV Pancreatic Cancer	Active	FOLFIRINOX	Unknown
121047830	66	Female	African-American	Stage IV Pancreatic Cancer	Active	Gemzar/Abraxane	None

CTC Biomarkers In PDAC Patients

Representative CTC QIF Images – Subject 120785056

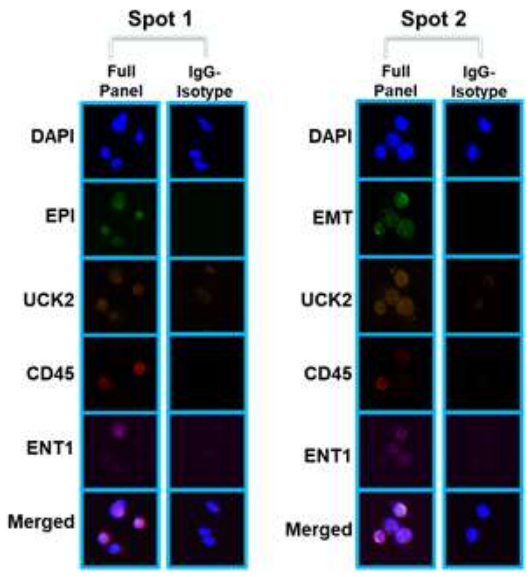
Spot 1
Full Panel
IgG-Isotype
DAPI
EPI
UCK2
CD45
ENT1
Merged
Spot 2
Full Panel
IgG-Isotype
DAPI
EMT
UCK2
CD45
ENT1
Merged

Acknowledgements

This work was supported by Rexahn Pharmaceuticals and conducted at ApoCell. Pilot PDAC samples were obtained under an IRB-approved protocol (ApoCell). The ongoing trial (NCT03189914) is being conducted by Rexahn Pharnaceuticals, Inc.

rexahn PHARMACEUTICALS

PRECISION apostream