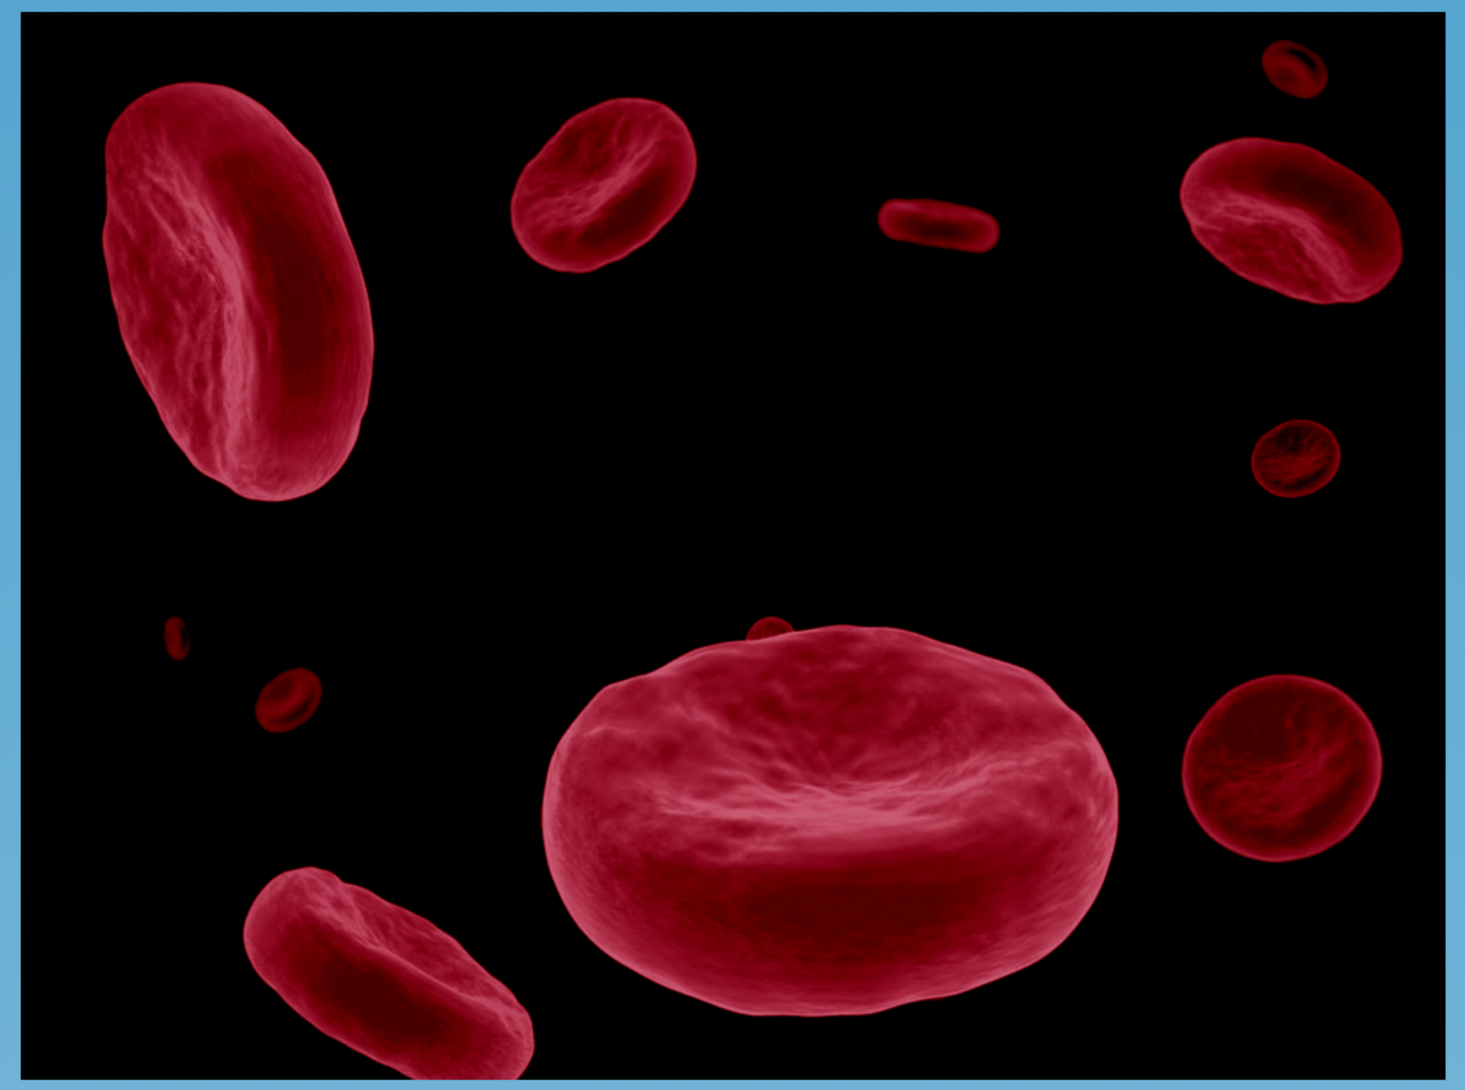

Single Molecule Dual JAK2-BET Inhibition as an MPN Therapeutic

Que T. Lambert1, Stuart W. Ember2, Muhammad Ayaz2, Harshani R. Lawrence2, Norbert Berndt2, Steven Gunawan2, Nicholas J. Lawrence2, Ernst Schönbrunn2 & **Gary W. Reuther**1

Departments of 1Molecular Oncology & 2Drug Discovery, Moffitt Cancer Center, Tampa, FL

MOFFITT CANCER CENTER

H. LEE MOFFITT CANCER CENTER & RESEARCH INSTITUTE, AN NCI COMPREHENSIVE CANCER CENTER – Tampa, FL
1-888-MOFFITT (1-888-663-3488) | MOFFITT.org

Panel 1: ABSTRACT

The optimism of anti-JAK2 therapy for the treatment of MPNs is largely based on the ability of JAK inhibitors to improve MPN patient constitutional symptoms. Unfortunately, such inhibitors are generally unable to induce remission or even allele burden, suggesting anti-JAK2 based therapies need refinement and optimization. Combination therapies continue to be widely investigated, however, such approaches could have significant complications when translating to patients. These include the difficulty determining proper dosing of each drug in combination in patients, the cost of treatment, and issues with combining drugs developed by different pharmaceutical companies.

Recently, we and others have identified anti-BET bromodomain inhibitory properties of various kinase inhibitors, including the JAK2 inhibitors TG101209 and TG101308. Bromodomains are protein-protein interaction motifs that bind to acetylated lysine and, for example, can play roles in regulating gene expression by binding acetylated histones. These dual kinase-BET inhibitors bind to the acetylated lysine-binding pocket of BET bromodomains and thus inhibit the function of such domains. Importantly, cancer cells appear to be particularly sensitive to BET inhibitors compared to normal cells. Combining a BET inhibitor and a JAK2 inhibitor has been shown to be more effective against MPN cells than JAK2 inhibition alone.

Our recent identification of dual kinase-BET inhibitors allows for the rational design of drugs with polypharmacology to inhibit more than one class of targets. To this end, we have optimized small molecules for dual anti-JAK2 and anti-BET activity. MA2-014 is a chemical derivative of TG101209 that exhibits similar anti-JAK2 activity, but about ten-fold improved anti-BET activity than TG101209. In fact, the activity of MA2-014 to target BET domains is similar to the prototypical BET inhibitor JQ1. For example, the IC50s of MA2-014 and JQ1 against the second bromodomain of the BET family member BRD4 are each about 20 nM. MA2-014 retains comparable biochemical activity against JAK2 as TG101209 and ruxolitinib (IC50s of low single digit nM, ~0.5 to 3 nM). In MPN cells, however, the ability of MA2-014 to inhibit JAK2-V617F signaling in MPN cells, as measured by P-STAT5, is about ten-fold improved over TG101209, and is comparable to ruxolitinib. Likewise, the ability of MA2-014 to inhibit the expression of c-Myc, which is widely used as a biomarker for BET inhibition, is about ten-fold better than TG101209 in MPN cells. The IC50 for MA2-014 for growth of Uke1 MPN cells is about 200 nM, compared to 500 nM for TG101209. Taken together, these data suggest that MA2-014 is a dual JAK2-BET inhibitor that exhibits superior BET inhibitory activity with similar if not better cellular JAK2 inhibitory activity than TG101209. MA2-014 efficiently inhibited the erythropoietin independent erythroid colony formation of myeloid progenitors from MPN patients, which is a hallmark of these cells and is widely used to test MPN therapeutics. The IC50 of MA2-014 in this assay is 50 nM, essentially identical to ruxolitinib, the only FDA approved JAK2 inhibitor for MPNs. The IC50 of TG101209 to inhibit this colony formation of primary MPN cells is 200 nM, four times greater than MA2-014. Interestingly, JAK2-V617F-driven MPN model cells that are resistant to ruxolitinib retained sensitivity to MA2-014. The IC50s of ruxolitinib and MA2-014 against the growth of BaF3-JAK2-V617F cells are about 100 nM. However, the same cells that are resistant to ruxolitinib (IC50 >2000 nM) remain sensitive to MA2-014 (IC50 of 240 nM). Finally, in long-term culture assays, we have determined that JAK2-V617F driven MPN Uke1 cells are not able to become resistant to MA2-014 as readily as they do to TG101209 or ruxolitinib. These data suggest MA2-014 may be more resilient to drug resistance, the major hurdle in the clinical effectiveness of JAK2 inhibitors.

Collectively, our work demonstrates that rationally-designed polypharmacology may be a novel approach to develop effective therapeutics for cancer, especially diseases that are driven by aberrant kinase signaling and are also sensitive to BET inhibition, as exemplified by MPNs. The use of such an optimized polypharmacologic therapeutic may provide the benefits of combination therapy with fewer complications associated with clinical development.

Panel 2: Combination of a BET Inhibitor and a JAK Inhibitor is Superior to Mono-therapy with Single Agents in MPN Cells.

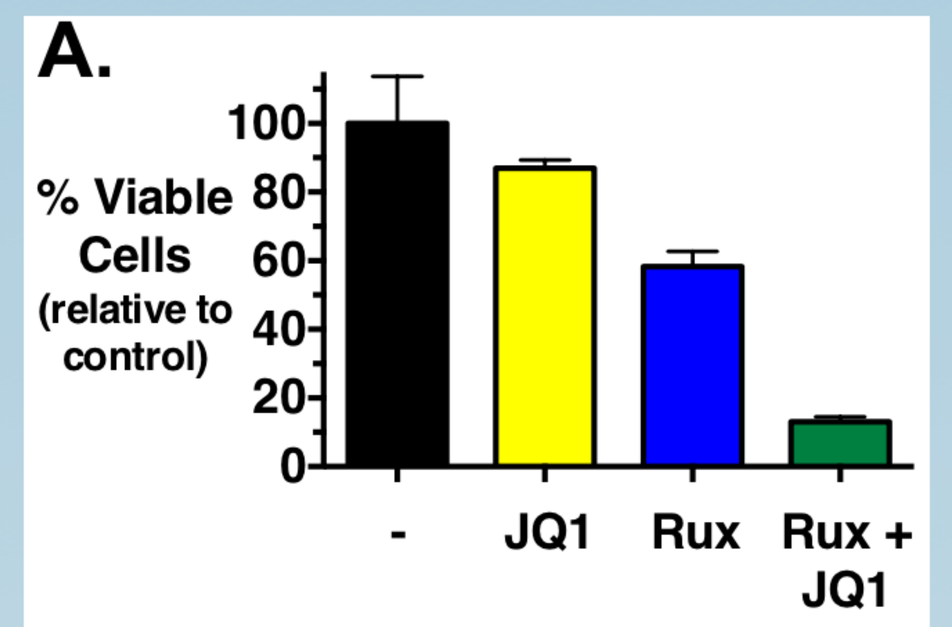

A.
% Viable Cells (relative to control)
100
80
60
40
20
0
-
JQ1
Rux
Rux + JQ1

B.

Drug	% Inhibition Observed	% Inhibition Expected if Additive
JQ1	13.1	
Rux	41.7	
Rux+JQ1	86.9	49.3

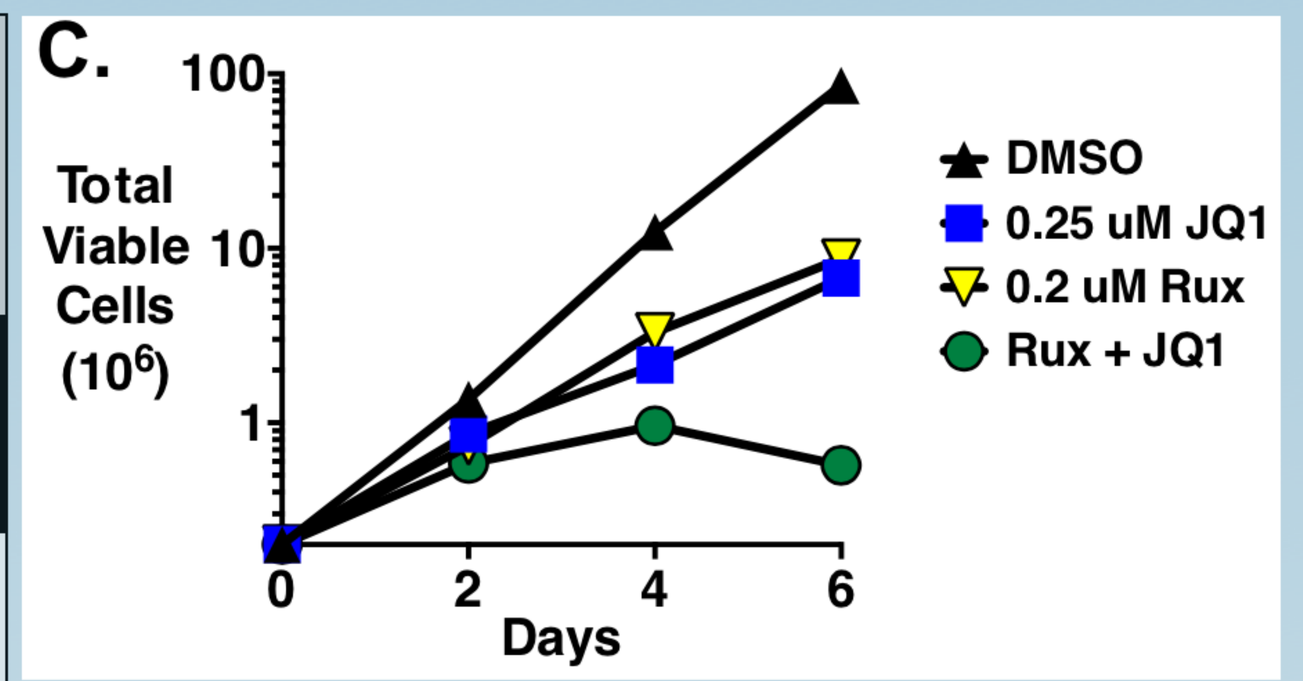

C.
Total Viable Cells (10^6)
100
10
1
0
2
4
6
Days
DMSO
0.25 uM JQ1
0.2 uM Rux
Rux + JQ1

Panel 2: Combination of a BET Inhibitor and a JAK Inhibitor is Superior to Mono-therapy with Single Agents in MPN Cells. A. HEL cells, which are patient-derived JAK2-V617F expressing cells that are sensitive to JAK2 inhibitors, were treated with DMSO as a control (black bar) & suboptimal concentrations (i.e. ≤ IC50) of JQ1 (0.3 uM, yellow bar), ruxolitinib (0.5 uM blue bar), or a combination of those concentrations of JQ1 & ruxolitinib (green bar). Relative total viable cells were determined by MTS assay after three days of culture & data was normalized to the DMSO sample. Error bars indicate s.d. **B.** Table showing observed % growth inhibition in A. for each treatment. The ruxolitinib & JQ1 combination treatment gave about 87% growth inhibition, which is greater than the expected additive effect of 49% (Bliss model). **C.** Uke1 cells, which are patient-derived JAK2-V617F expressing cells that are dependent on this JAK2 mutant for growth & viability, were treated as indicated. Total viable cells were determined over time by trypan blue exclusion.

Development of an Optimized Dual JAK2-BET Inhibitor, MA2-014

Panel 3: TG101209 Co-crystal Structures with JAK2 Kinase Domain and BRD4 BET Domain.

JAK2:TG101209

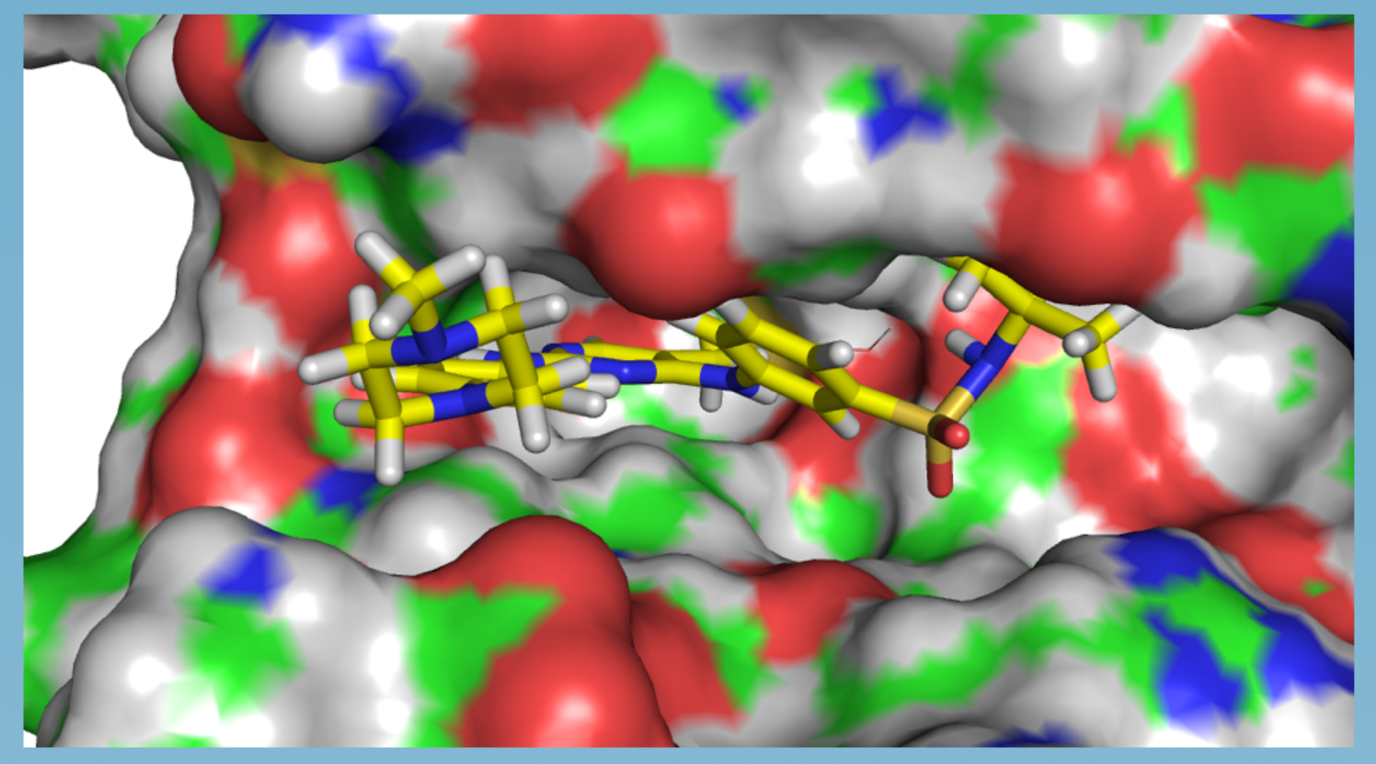

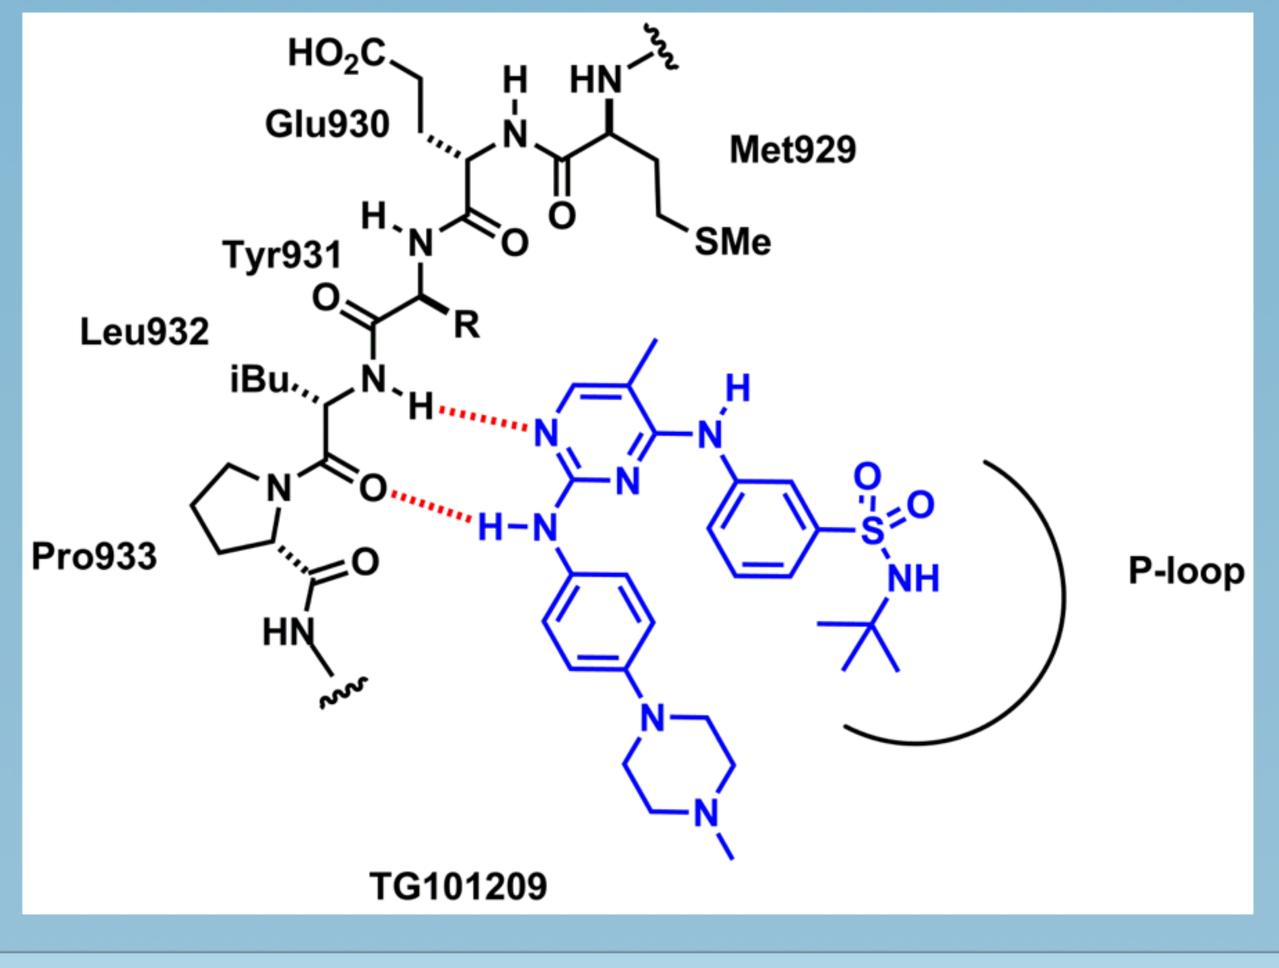

HO_2C
Glu930
Met929
SMe
Tyr931
Leu932
iBu
R
Pro933
P-loop
NH
TG101209

BRD4 BET:TG101209

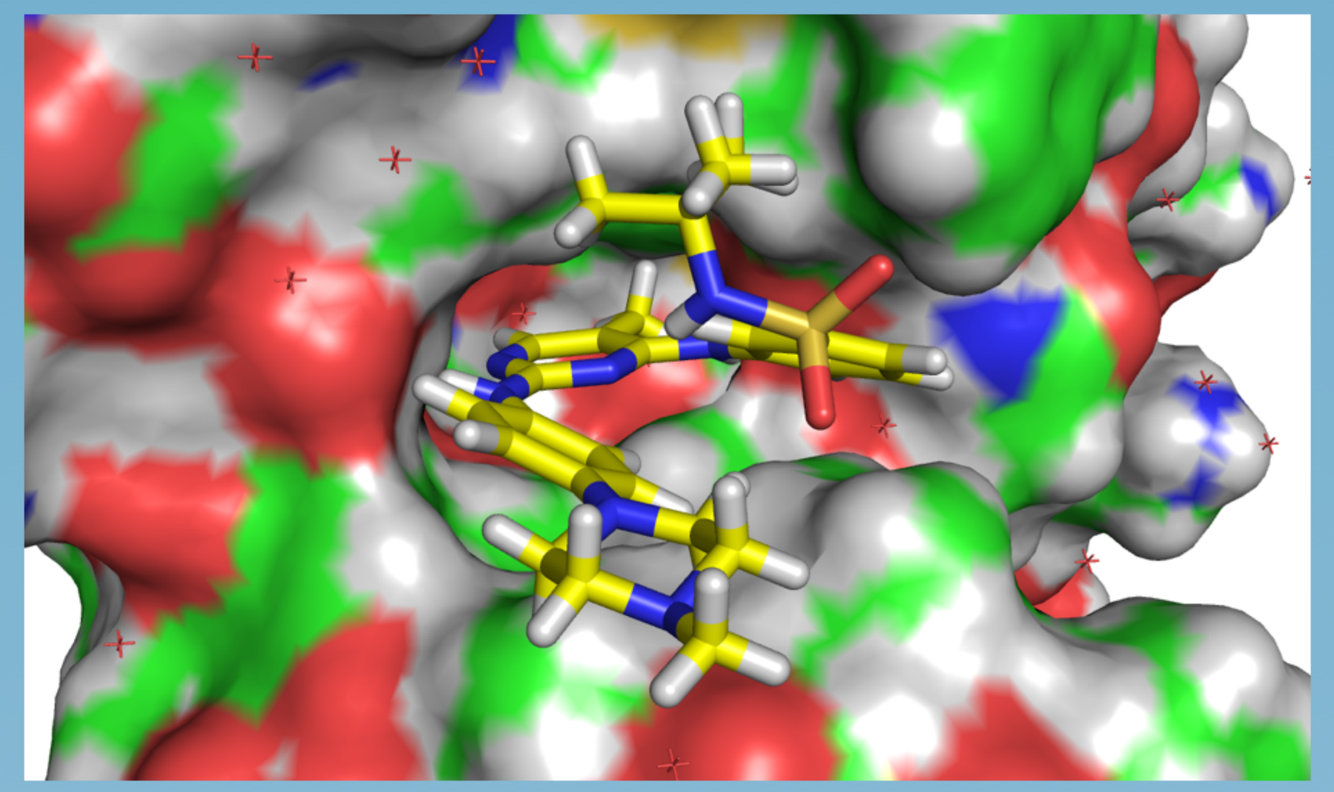

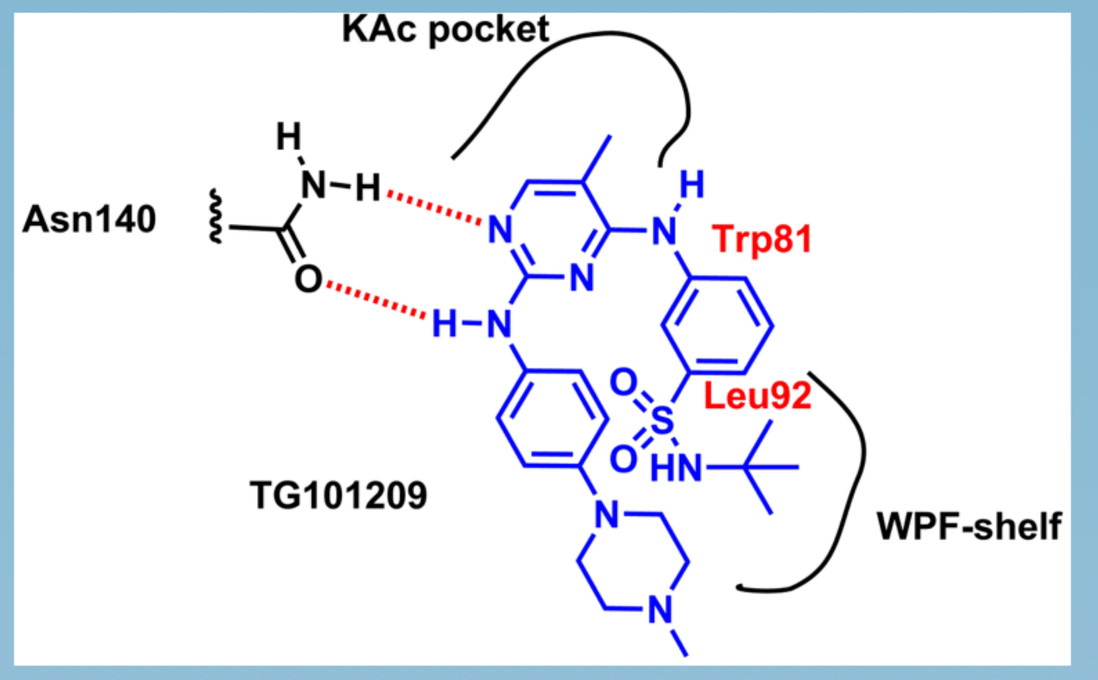

KAc pocket
Asn140
Trp81
Leu92
TG101209
WPF-shelf

Panel 4: MA2-014 is an Optimized Dual JAK2-BET Inhibitor Analog of TG101209. The BET binding activity of MA2-014 is ten-fold improved over that of TG101209.

IC50s of Compounds Against JAK2 and the First Bromo/BET Domain of BRD4

Compound	IC50 against JAK2 (nM)	Binding potential for BRD4-1 bromodomain (IC50, nM)
TG101209	0.5	113
MA2-014	1.04	10.2
Ruxolitinib	2.8 ref 60	not a BET inhibitor *
JQ1	not a JAK2 inhibitor **	22.2

IC50s of Compounds against BET Domains and Kinases

	Target IC50 (nM)								
Compound	BRD2-1	BRD3-1	BRD4-1	BRDT-1	BRD4-2	JAK1	JAK2	JAK3	FLT3
MA2-014	625	12.5	10.2	29.1	23.6	4.58	1.04	91.4	0.917
TG101209	680	130	113	290	172	2.63	0.508	97.6	1.53
JQ1	99.7	16.5	22.2	159	19.7	not active *			n.d.

Panel 4: IC50s of Compounds against BET Domains and Kinases

- *Top table*: Compounds were screened against JAK2 and the first bromodomain of BRD4.
- *Bottom table*: Compounds were screened against the indicated BET domains (e.g. BRD4-1 indicates the first BET domain of BRD4) & kinases using AlphaScreen Assay for BETs and 32P-labelled kinase reactions (HotSpot Kinase Assay) for kinases (by Reaction Biology Corp.).

Red = ~Ten-fold improvement over TG101209 & in most cases similar to JQ1.

JQ1 is not a JAK inhibitor (* Liu et al. 2014 Mol Cancer Ther. 13:1194. & Panel 5) and ruxolitinib does not inhibit BET (** Ciceri et al. Nat Chem Biol. 10:305).
n.d., not determined.

Panel 5: Effect of MA2-014, a Novel Dual JAK2-BET inhibitor, on MPN Cells.

Cell Line	IC50 (nM) MA2-014	IC50 (nM) TG101209
Uke1 (MPN, JAK2 driven)	195	550
Set2 (MPN, JAK2 driven)	209	not determined
JZ531-1 (MPN, JAK2 driven)	252	
BaF3/JAK2-V617F (JAK2 driven)	100	
Jurkat (non MPN, not JAK2- driven)	1900	

- *Table above*: The IC50s, as determined by MTS assay, of MA2-014 on JAK2-V617F-dependent MPN cell lines & Jurkat cells as a non-JAK2-dependent control, & TG101209 on Uke1 cells are shown.
- *Blots to right*: **Improved Dual JAK2-BET Inhibition in Cells.** The dual JAK2-BET inhibitor MA2-014 (top blot) more effectively inhibits both JAK2 activity (as indicated by pSTAT5) & BET (indicated by c-Myc expression) than TG101209 in 6-hr treatment of Uke1 cells. The JAK2 inhibitor ruxolitinib, which is not a BET inhibitor, inhibited JAK2 but only had a modest non-dose dependent effect on c-Myc levels. The BET inhibitor JQ1 (bottom blot) decreased c-Myc levels but did not inhibit JAK2 (pSTAT5) as expected (Liu et al. 2014 Mol Cancer Ther. 13:1194).

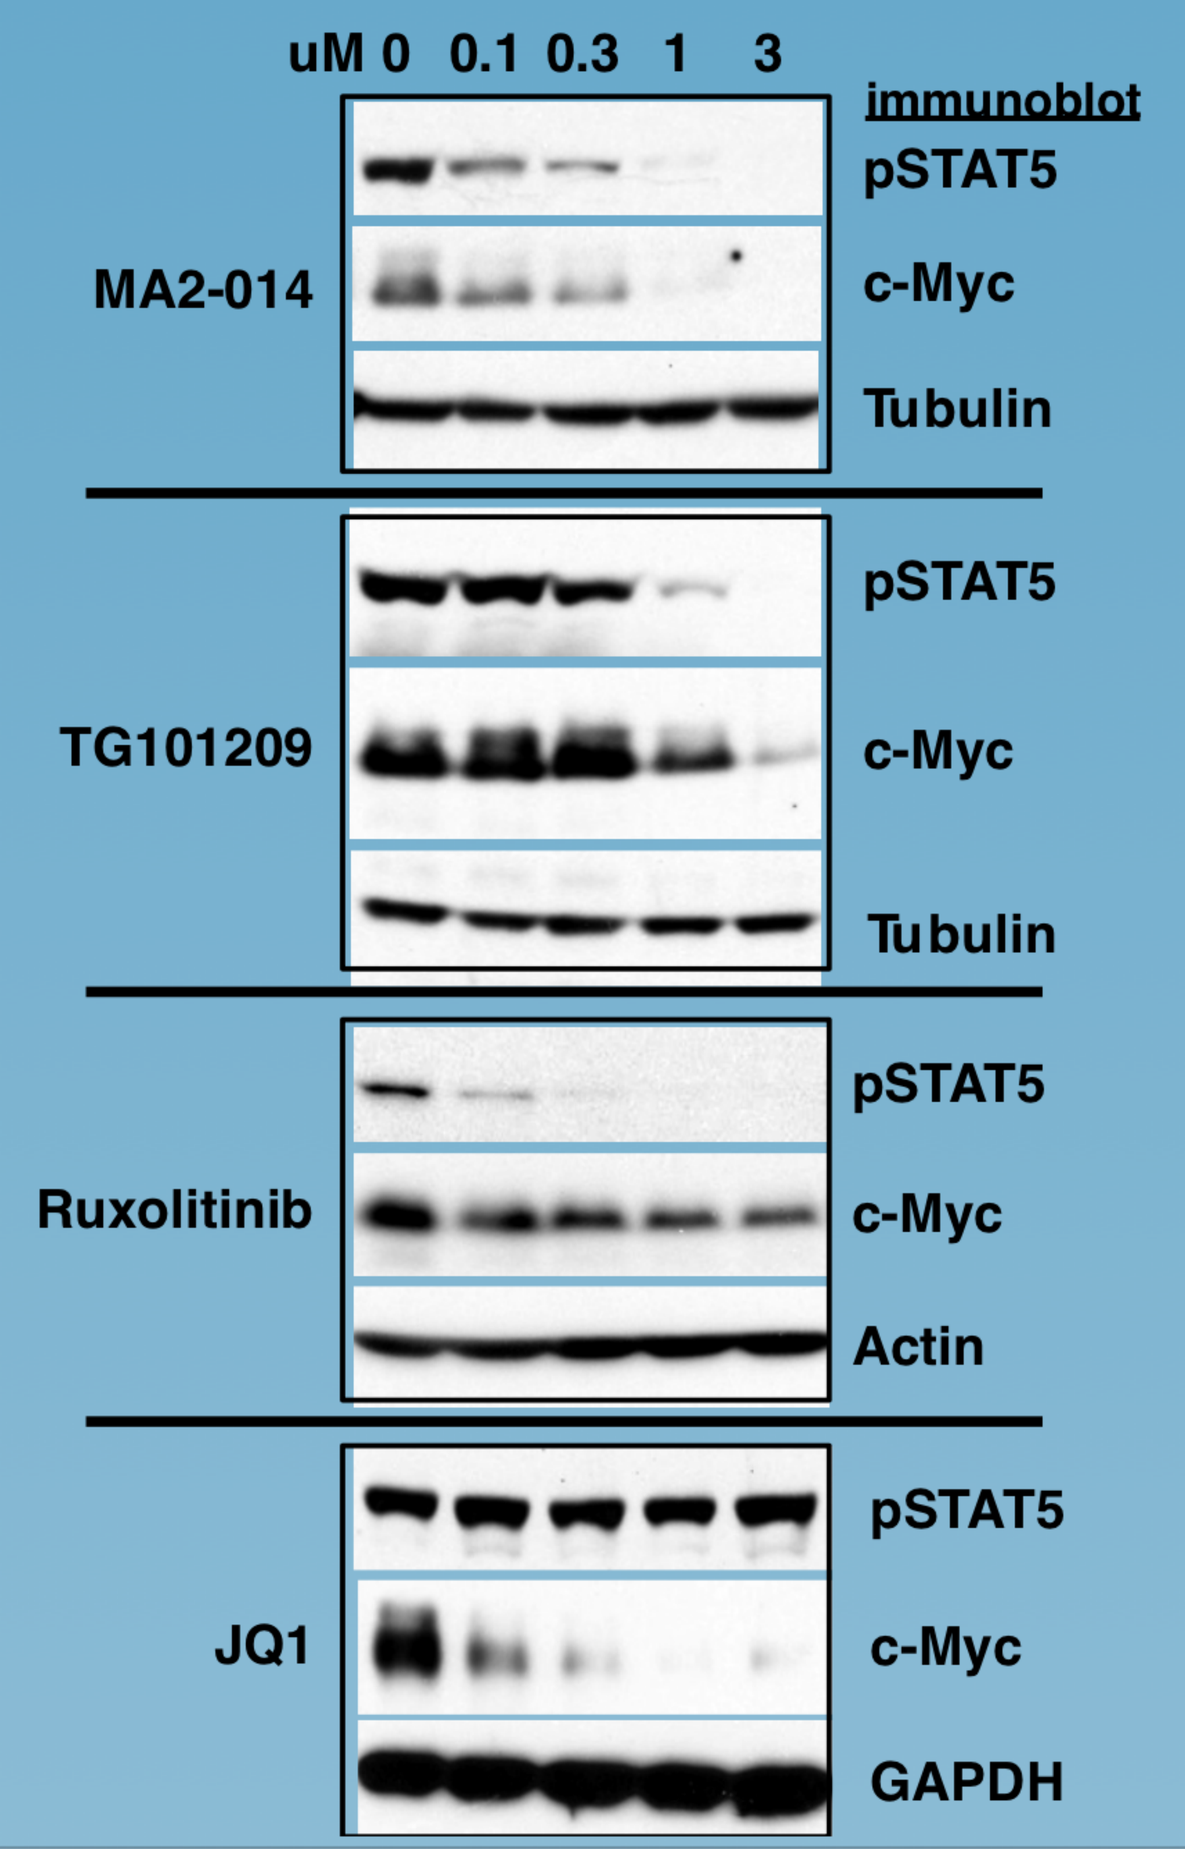

uM 0 0.1 0.3 1 3
immunoblot
MA2-014
pSTAT5
c-Myc
Tubulin
TG101209
pSTAT5
c-Myc
Tubulin
Ruxolitinib
pSTAT5
c-Myc
Actin
JQ1
pSTAT5
c-Myc
GAPDH

Panel 6: MA2-014 is More Efficacious Than TG101209 at Inhibiting Erythroid Colony Formation of Cells from MPN Patients.

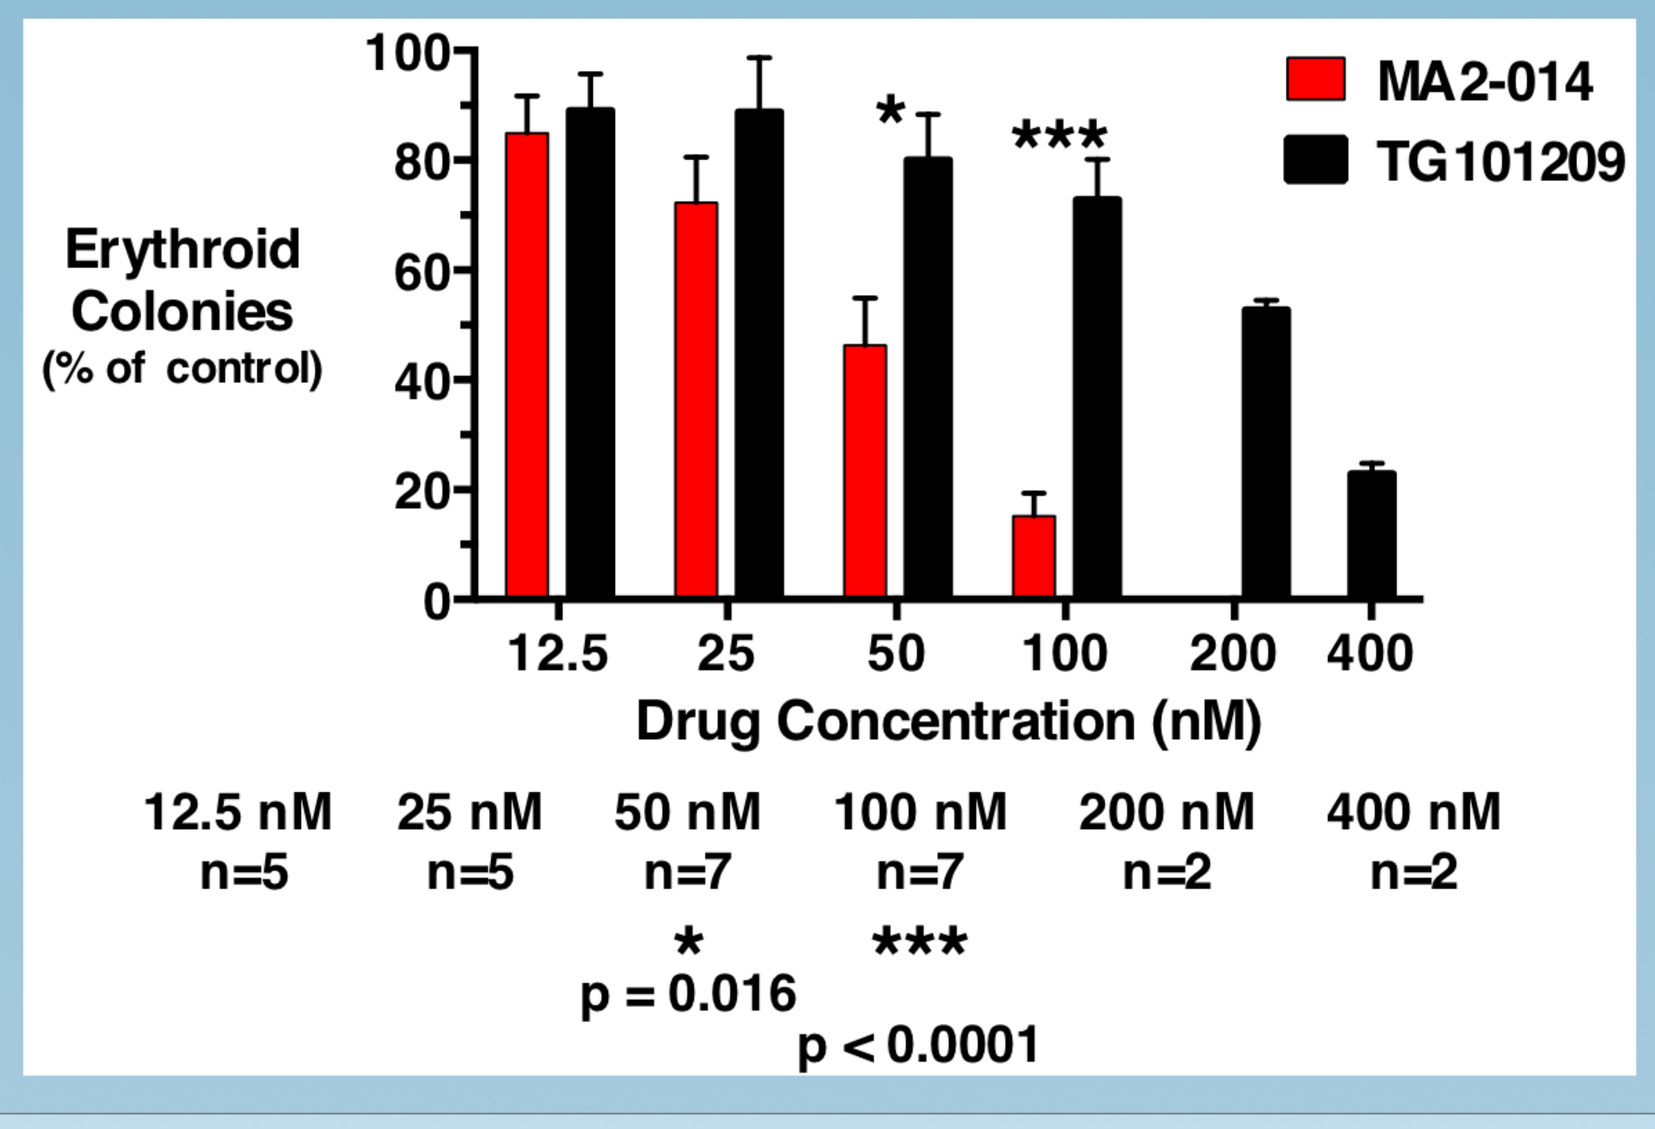

Erythroid Colonies (% of control)
100
80
60
40
20
0
MA2-014
TG101209
*

12.5
25
50
100
200
400
Drug Concentration (nM)
12.5 nM n=5
25 nM n=5
50 nM n=7
100 nM n=7
200 nM n=2
400 nM n=2
* p = 0.016
*** p < 0.0001

Peripheral blood mononuclear cells of JAK2-V617F-positive MPN patients were plated in methylcellulose containing cytokines but no erythropoietin, with the indicated concentrations of MA2-014 and TG101209. Erythroid colonies (erythropoietin-independent) were counted after 14 days and the number of colonies is represented as a percent of the number of colonies obtained with untreated (DMSO) cells. Error bars indicate standard error of the indicated sample numbers. p values calculated by t-test.

Panel 7: JAK2 Inhibitor Cells Remain Sensitive to MA2-014 and MPN Cells are Less Able to Develop Resistance to MA2-014 than to TG101209 or to Ruxolitinib.

	IC50 (nM)	
Drug	***Sensitive***	***Resistant***
ruxolitinib	120	> 2500
TG101209	420	2000
MA2-014	140	240

Table above: Both JAK2 inhibitor sensitive & resistant BaF3-JAK2-V617F cells were treated with ruxolitinib, TG101209, & MA2-014. While resistant cells were highly resistant to ruxolitinib & TG101209, these cells remained sensitive (red font) to MA2-014. IC50s (MTS assays) are shown.
Graph to right: Uke1 cells (1x105/mL) were cultured in DMSO, MA2-014, ruxolitinib, or TG101209, each at 50 nM. Every 2-3 days cells were counted by trypan blue exclusion & when cells demonstrated resistance (>= 90% growth of DMSO control), the drug concentration was doubled. Data is presented as drug concentration the cells were cultured in over time & therefore drug resistance is directly relative to the increasing slope of the data.

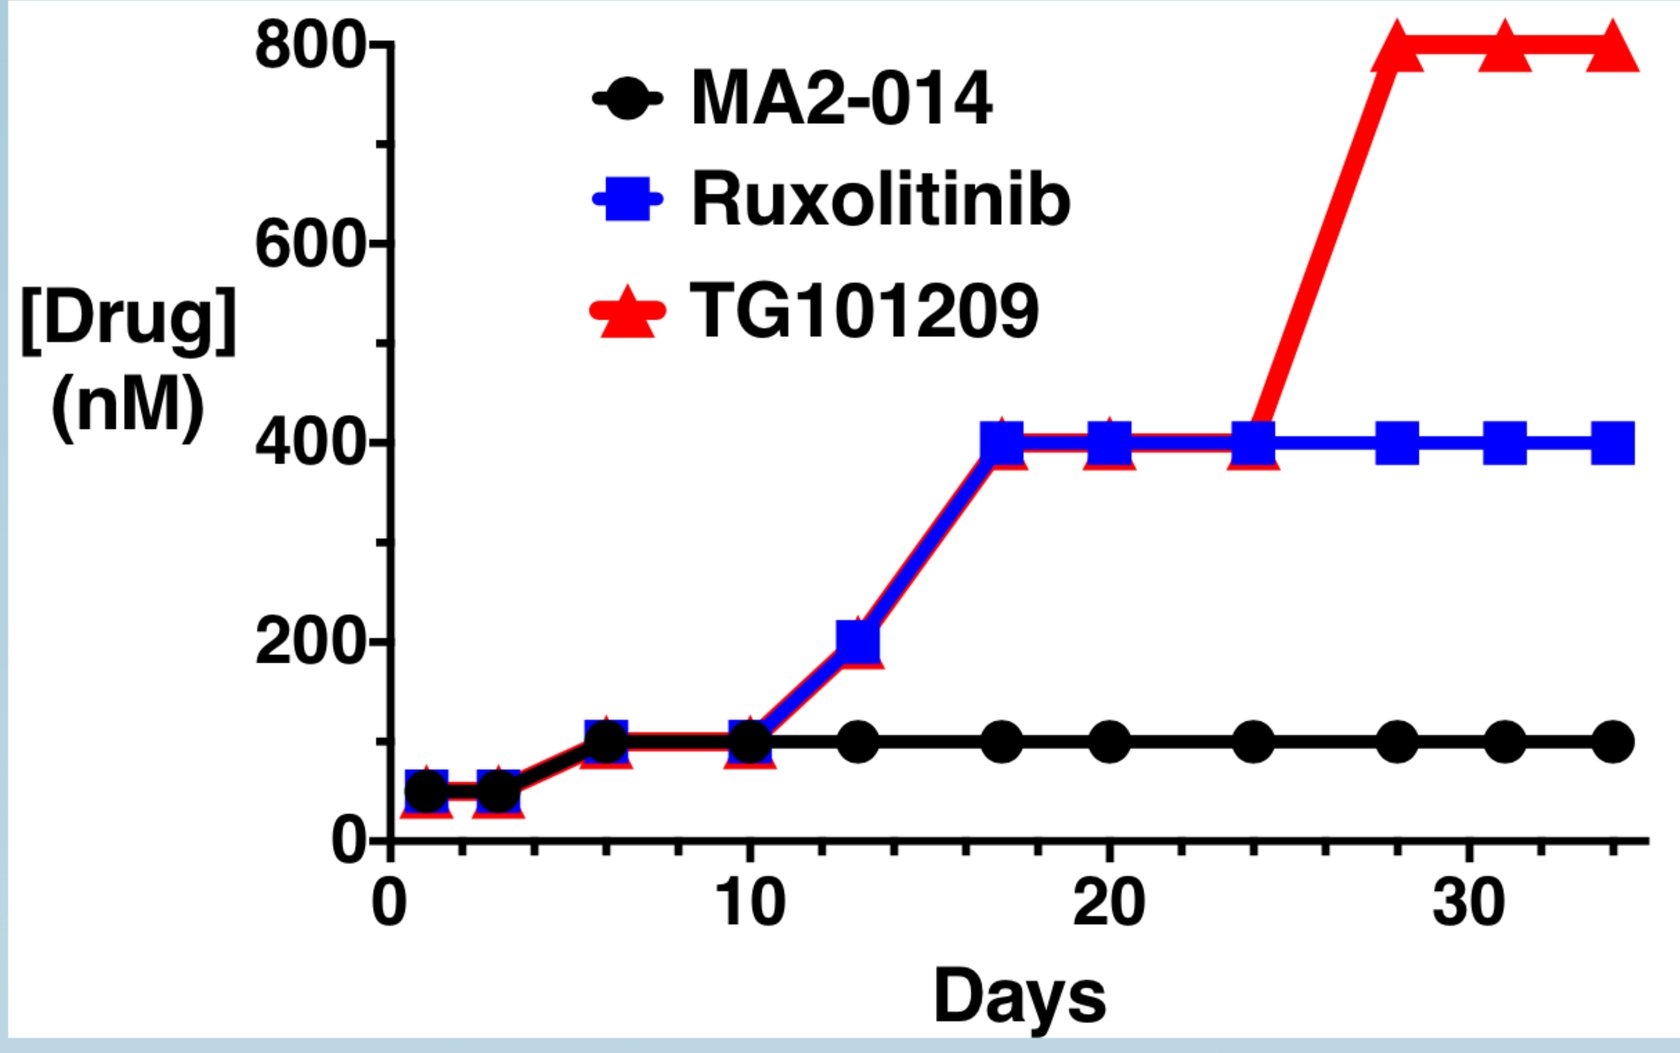

[Drug] (nM)
800
600
400
200
0
0
10
20
30
Days
MA2-014
Ruxolitinib
TG101209

SUMMARY

1. MA2-014 is a JAK2-BET inhibitor developed using structural analyses of TG101209 w/ JAK2 & BRD4 BET domain.
2. MA2-014 has 10-fold improved BET inhibitory activity than TG101209 while retaining potent anti-JAK2 activity.
3. MA2-014 exhibits superior growth inhibition of MPN cells, including cell lines & primary cells from MPN patients.
4. Initial studies suggest JAK2 inhibitor resistant cells remain sensitive to the dual JAK2-BET inhibitor MA2-014 and MPN cells can not develop resistance to MA2-014 as readily as to ruxolitinib.
5. Single molecule dual inhibitors, such as the dual JAK2-BET inhibitor MA2-014, may overcome combination therapy-associated obstacles & improve MPN therapeutics, perhaps by thwarting drug resistance development.

The candidates included in these studies, along with all related therapeutic candidates from Moffitt Cancer Center inhibiting bromodomain family proteins, have been licensed to Aptose Biosciences Inc.

There are no relevant conflicts of interest to disclose.