Effect of SCY-078 on the Pharmacokinetics of a CYP2C8 substrate (Rosiglitazone)- Results from a Phase 1 Clinical Trial

27th ECCMID
Vienna, Austria
22 – 25 April 2017

Angulo David [1], Murphy Gail [2], Atiee George [3], Corr Christy [4], Willett Michael [5], Wring Steve [1]
1. SCYNEXIS, Inc. NJ, US; 2. Riverside Consulting, DE, US; 3. Worldwide Clinical Trials TX, USA; 4. BCH Research Solutions PA, US; 5. Ready Clinical NJ, US.

INTRODUCTION & PURPOSE

- SCY-078 is an oral and intravenous semi-synthetic triterpenoid antifungal glucan synthase inhibitor, in development for the treatment of invasive and mucocutaneous fungal diseases.

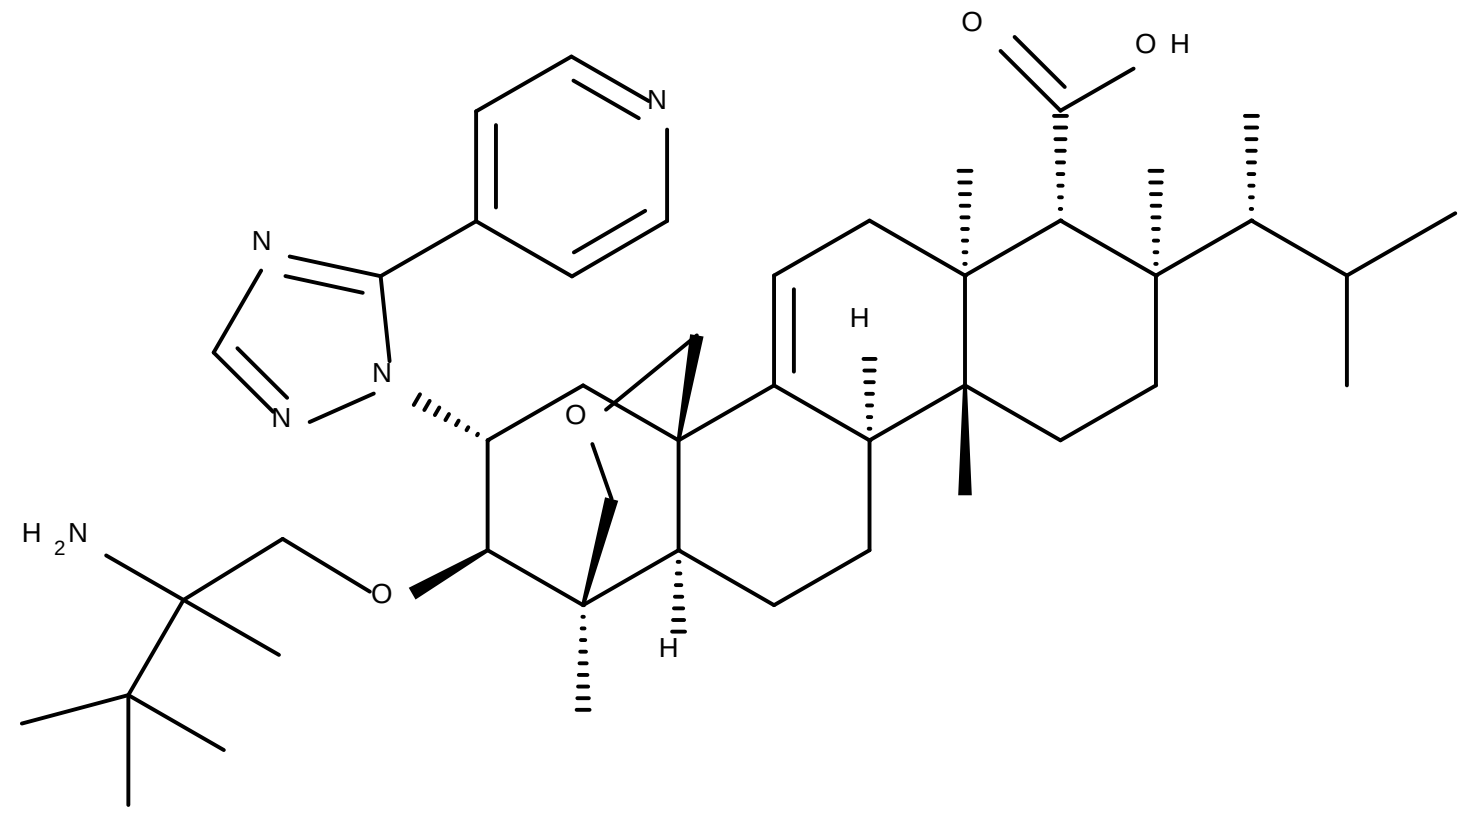

- *In vitro* data show that SCY-078 has potential to inhibit CYP2C8 and an ~5x lower potential to inhibit CYP3A; it does not inhibit other CYP isozymes.
- We conducted a clinical drug-drug interaction trial to characterize any potential effects of SCY-078 co-administered to steady-state on the pharmacokinetics of rosiglitazone, a medication that is sensitive to inhibition of CYP2C8, as an indicator of the potential for clinically meaningful interaction with drugs metabolized via the CYP family of enzymes.

METHODS: STUDY DESIGN

Phase 1, open-label, randomized, 2-period crossover study consisting of two treatments administered in random order separated by a minimum 10-day washout interval between periods in 24 healthy adult male or female subjects. Subjects received two oral treatments in random sequence:

- Treatment A = a single oral 4-mg dose of rosiglitazone given alone on Day 1
- Treatment B = Oral 1250-mg SCY-078 on Day 1 (loading) followed by a QD dose of oral 750-mg SCY-078 for 7 days with a single 4-mg dose of rosiglitazone administered concurrently on Day 3 (predicted to be at steady-state exposure for SCY-078).

Safety was monitored throughout the study by repeated clinical and laboratory evaluations.
Plasma samples were obtained for determination of SCY-078, rosiglitazone and N-desmethyl rosiglitazone (rosiglitazone's metabolite) concentrations.

CONCLUSION

There is no clinically meaningful impact on rosiglitazone plasma levels when co-administered with steady-state SCY-078, supporting that SCY078 is not an inhibitor of CYP2C8 or other CYP isozymes where the inhibitory potency of SCY-078 is even lower.

RESULTS

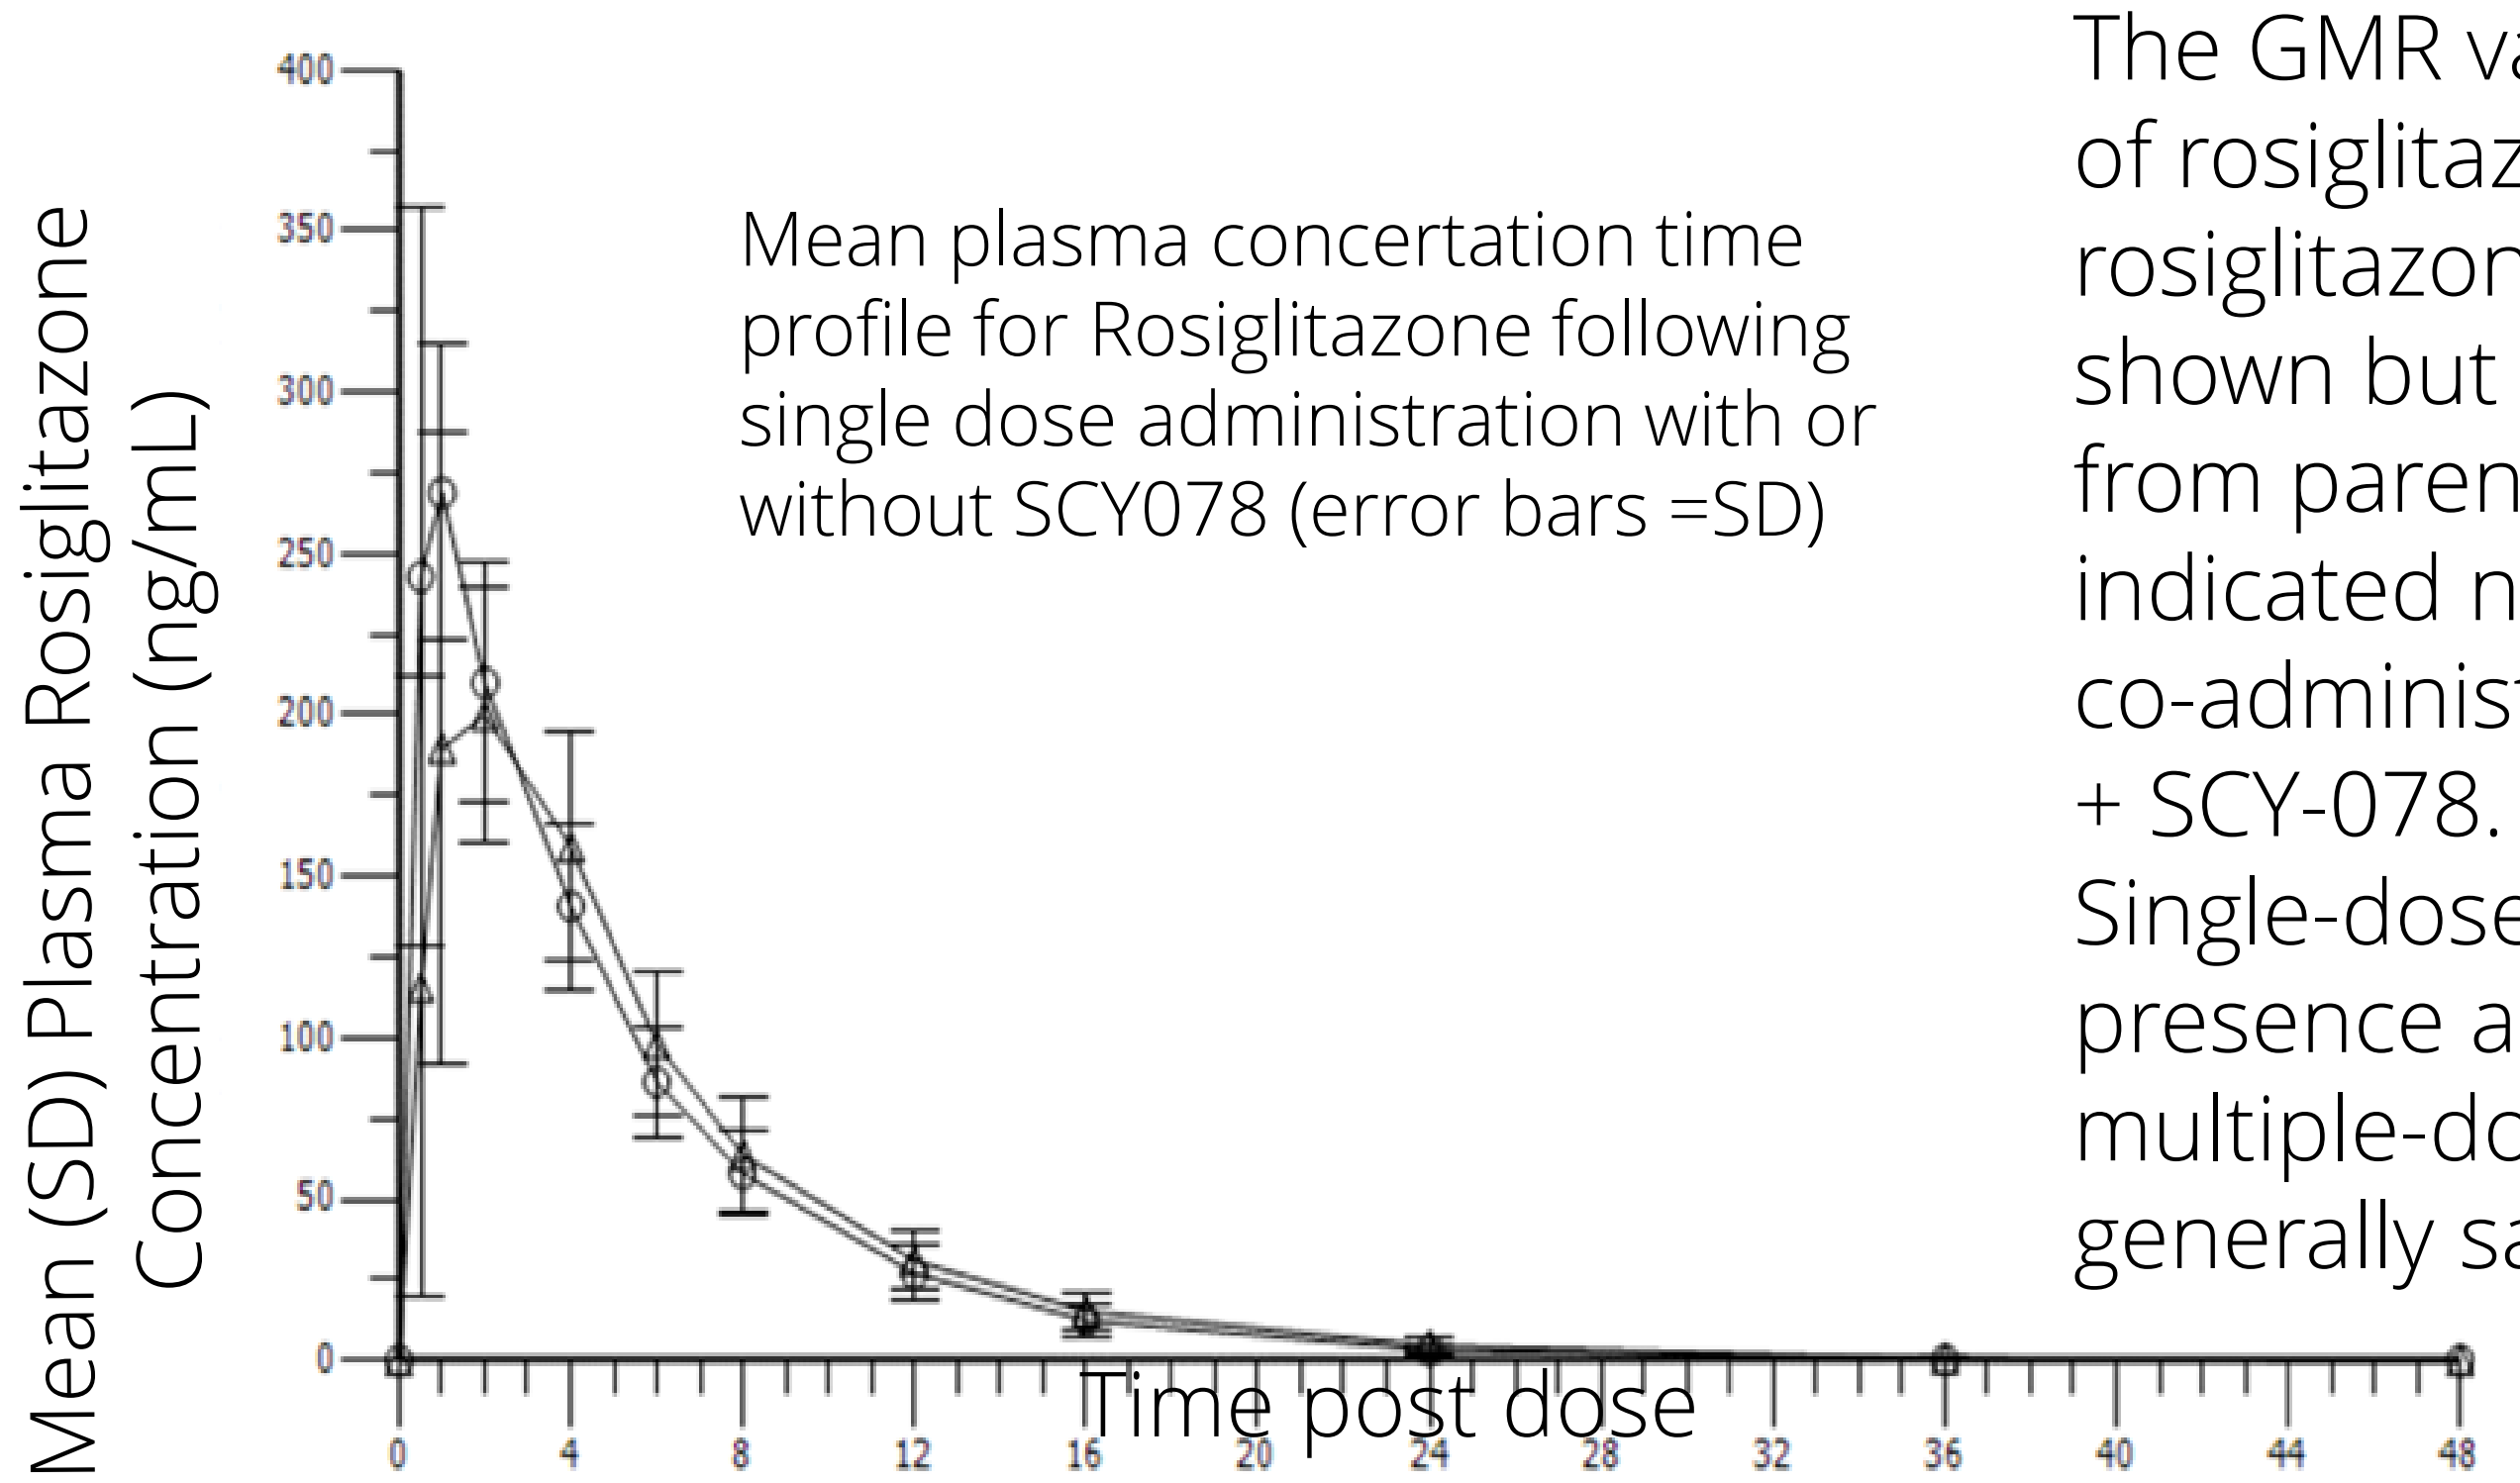
Mean (SD) Plasma Rosiglitazone Concentration (ng/mL)
400
350
300
250
200
150
100
50
0
Time post dose
0
4
8
12
16
20
24
28
32
36
40
44
48

Mean plasma concertation time profile for Rosiglitazone following single dose administration with or without SCY078 (error bars =SD)

The GMR values for the analysis of rosiglitazone and N-desmethyl rosiglitazone (metabolite data not shown but consistent with data from parent rosiglitazone) indicated no relevant effect of the co-administration of rosiglitazone + SCY-078.
Single-dose rosiglitazone in the presence and absence of oral multiple-doses of SCY 078 were generally safe and well-tolerated.

PK Rosiglitazone Co-administered with SCY-078 vs. Rosiglitazone Alone

Treatment	AUC_{0-Inf} (h*nM)[a]	C_{max} (nM)[a]	T_{max} (h)[b]
Test (Rosiglitazone + SCY-078)	1451 (1334, 1578)	235.1 (211.9, 260.7)	1.0 (0.5 - 4.0)
Reference (Rosiglitazone Alone)	1461 (1344, 1589)	295.6 (266.5, 327.8)	0.53 (0.5 - 1.0)
GMR[c] Test/Reference (90% CI).	0.99 (0.90, 1.10)	0.80 (0.70, 0.90)	

[a]LS Geometric Mean and its 95% CI were calculated based on linear model: log(PK Result)=Sequence o f Treatment. ; [b]Median (Min-Max).; [c]Geometric Means Ratio, GMR

For additional information, contact us at info@scynexis.com.

P1713