Poster # 250

LENABASUM REDUCES LPS-INDUCED INFLAMMATION IN AIRWAY MACROPHAGES FROM HUMAN CYSTIC FIBROSIS LUNGS

Bob A. Lubamba[1], George Zhang[2], Mark Tepper[2], and Carla M. P. Ribeiro[1]

1. University of North Carolina, Chapel Hill, NC, USA; 2. Corbus Pharmaceuticals, Norwood, MA, USA.

Background

Cystic fibrosis (CF) airways exhibit excessive and persistent inflammation partly driven by airway macrophages (AMs)[1]. Lenabasum (fka anabasum) is an oral synthetic cannabinoid receptor type 2 (CB2) agonist that has been shown to reduce the production of key airway pro-inflammatory cytokines known to play a role in CF[2]. Lenabasum acts by activating the resolution of inflammation, an endogenous pathway that restores immunological homeostasis[3].

In a recently completed double blind placebo-controlled Phase 2 study with 85 adults diagnosed with CF, lenabasum reduced pulmonary exacerbations and decreased inflammatory cells and mediators in sputa while demonstrating a favorable safety profile[4]. The study recruited patients regardless of their CFTR mutation, their lung pathogens or their current standard of care (including approved CFTR-targeting medications).

In this study, we further evaluated lenabasum by testing its effects on AMs from harvested human CF lungs.

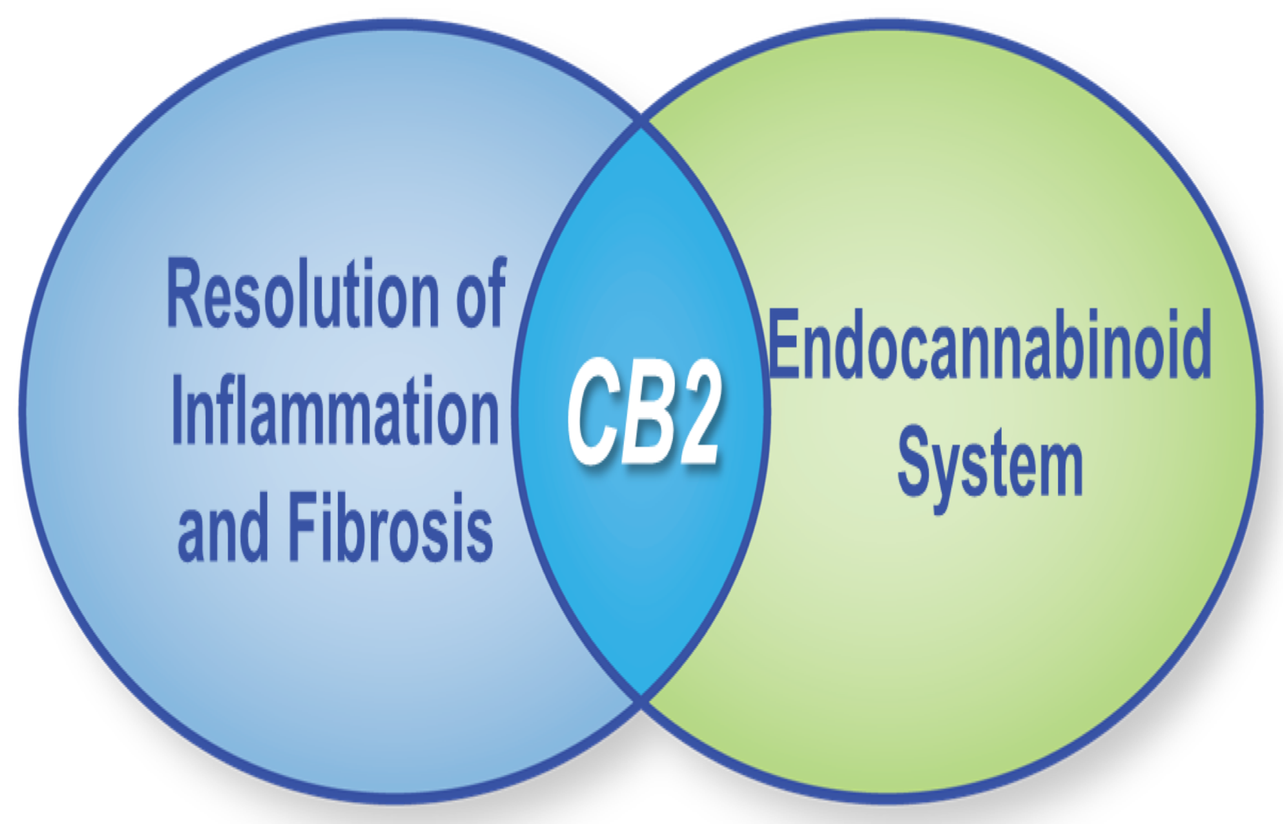
Resolution of Inflammation and Fibrosis
CB2
Endocannabinoid System

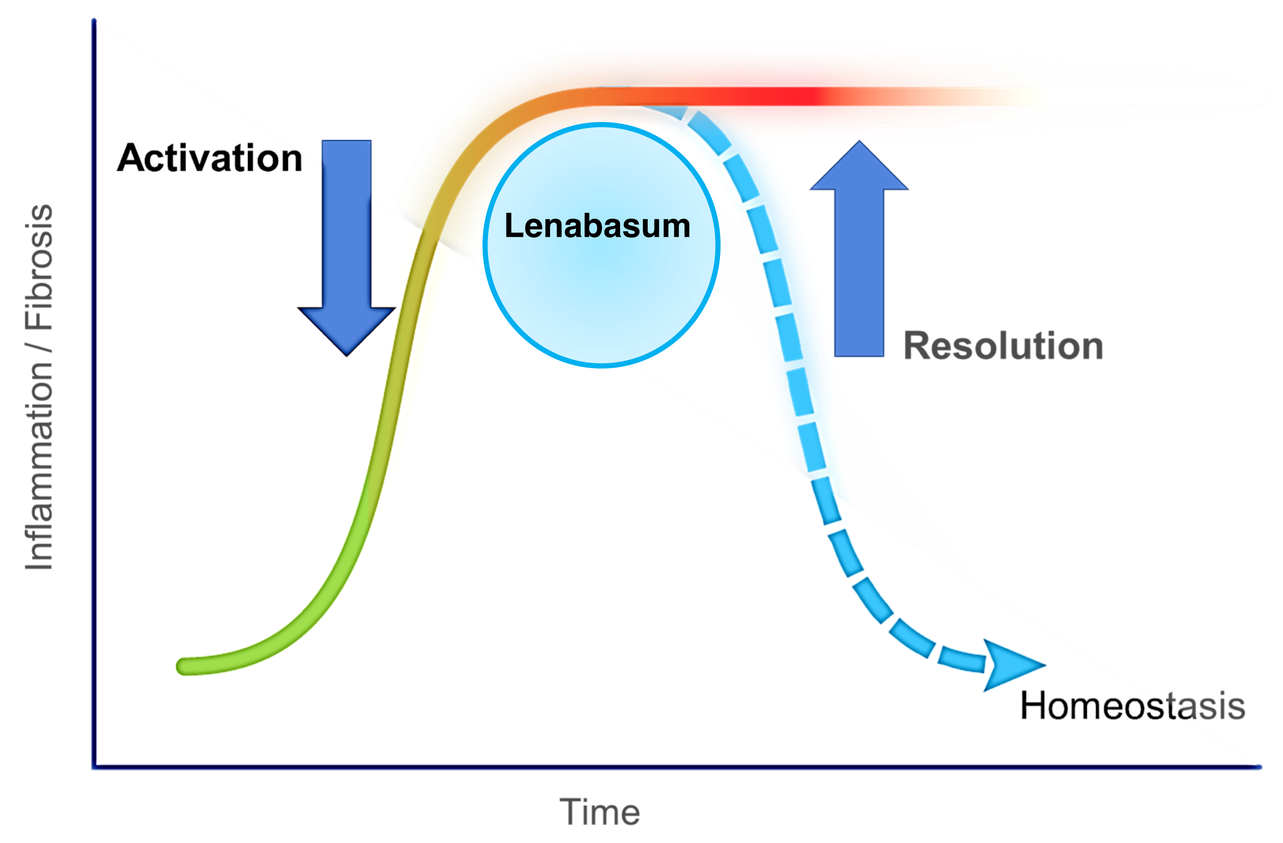
Activation
Lenabasum
Resolution
Homeostasis
Inflammation / Fibrosis
Time

Resolution of Chronic Inflammation and Fibrosis

Objective

To determine the effect of lenabasum on production and secretion of inflammatory cytokines by CF AMs.

Methods

AM Culture

AMs were recovered from surgically removed CF lungs as previously described[1]. A balloon-tipped catheter was placed into the largest available bronchus under aseptic conditions. The lungs were perfused with PBS and the retrieved fluid was centrifuged (250 x g for 10 min, 4°C). The cell pellet was resuspended in macrophage medium. Freshly isolated CF AMs were seeded onto 24-well plates at a concentration of 1×10^5 AMs per well and cultured in macrophage medium. The lungs were homozygote for F508del CFTR.

Treatment

Adherent AMs were washed with PBS and treated with 100 ng/ml LPS from *Pseudomonas aeruginosa* (Sigma-Aldrich, MO, USA). A treatment of 6 h was selected based on maximal mRNA transcript expression and detection of protein secretion in 3 day old cultured AMs. Treatment with vehicle (0.1% DMSO) did not affect the expression of inflammatory markers, as compared with naïve untreated AMs. Therefore, cytokine values obtained under 0.1% DMSO treatment were considered baseline values.

Lenabasum was used at the concentration of 1, 3 and 10 µM

ELISA assays

The secretion of TNF-α, IL-6, IL-8, LXA4 and PGJ2 was evaluated using commercially-available specific human ELISA kits (R&D System, Minneapolis, MN, USA), following the manufacturer's instructions.

RT-PCR

Harvested cells were washed in ice-cold PBS, and the total RNA was extracted using Trizol (Invitrogen, CA, USA). Total RNA concentration was measured by absorption at 260 nm, and purity and concentration were checked by determining the OD ratio 260/280nm using a NanoDrop-8000 spectrophotometer (Thermo Scientific, MA, USA). Total RNA was reversed transcribed into cDNA with iScriptTM cDNA Synthesis kit for quantitative real-time (qRT)-PCR (Bio-Rad, CA, USA). All samples were reverse transcribed under the same conditions. Quantitative gene expression and subsequent data analyses were performed using the Applied Biosystems® 7500 Real-Time PCR Systems (Applied Biosystems, Darmstadt, Germany). Briefly, duplicate reactions were performed using 2 μl first-strand cDNA template, 7 μl deionized water, 1μl of TaqMan® MGB probes and 10 μl iTaqTM Universal Probes Supermix (Bio-Rad, CA, USA) master mix. The thermal cycling conditions were 3 min at 95°C followed by 40 cycles of 15s at 95°C and 1 min at 60 °C. The mRNA copy numbers of the target and reference genes were calculated according to the standard curve method. The data obtained from the qRT-PCR analyses were expressed as mean ± SEM. Normalization of data was performed by calculating the ratio between the gene of interest and the reference gene (18S) according to the procedure. The following Taqman Probes (Applied Biosystems, CA, USA) were used to assess the mRNA levels of the following genes: 18S: Hs99999901_s1; SPHK1:Hs01116530_g1 and XBP-1s:Hs03929085_g1.

Results

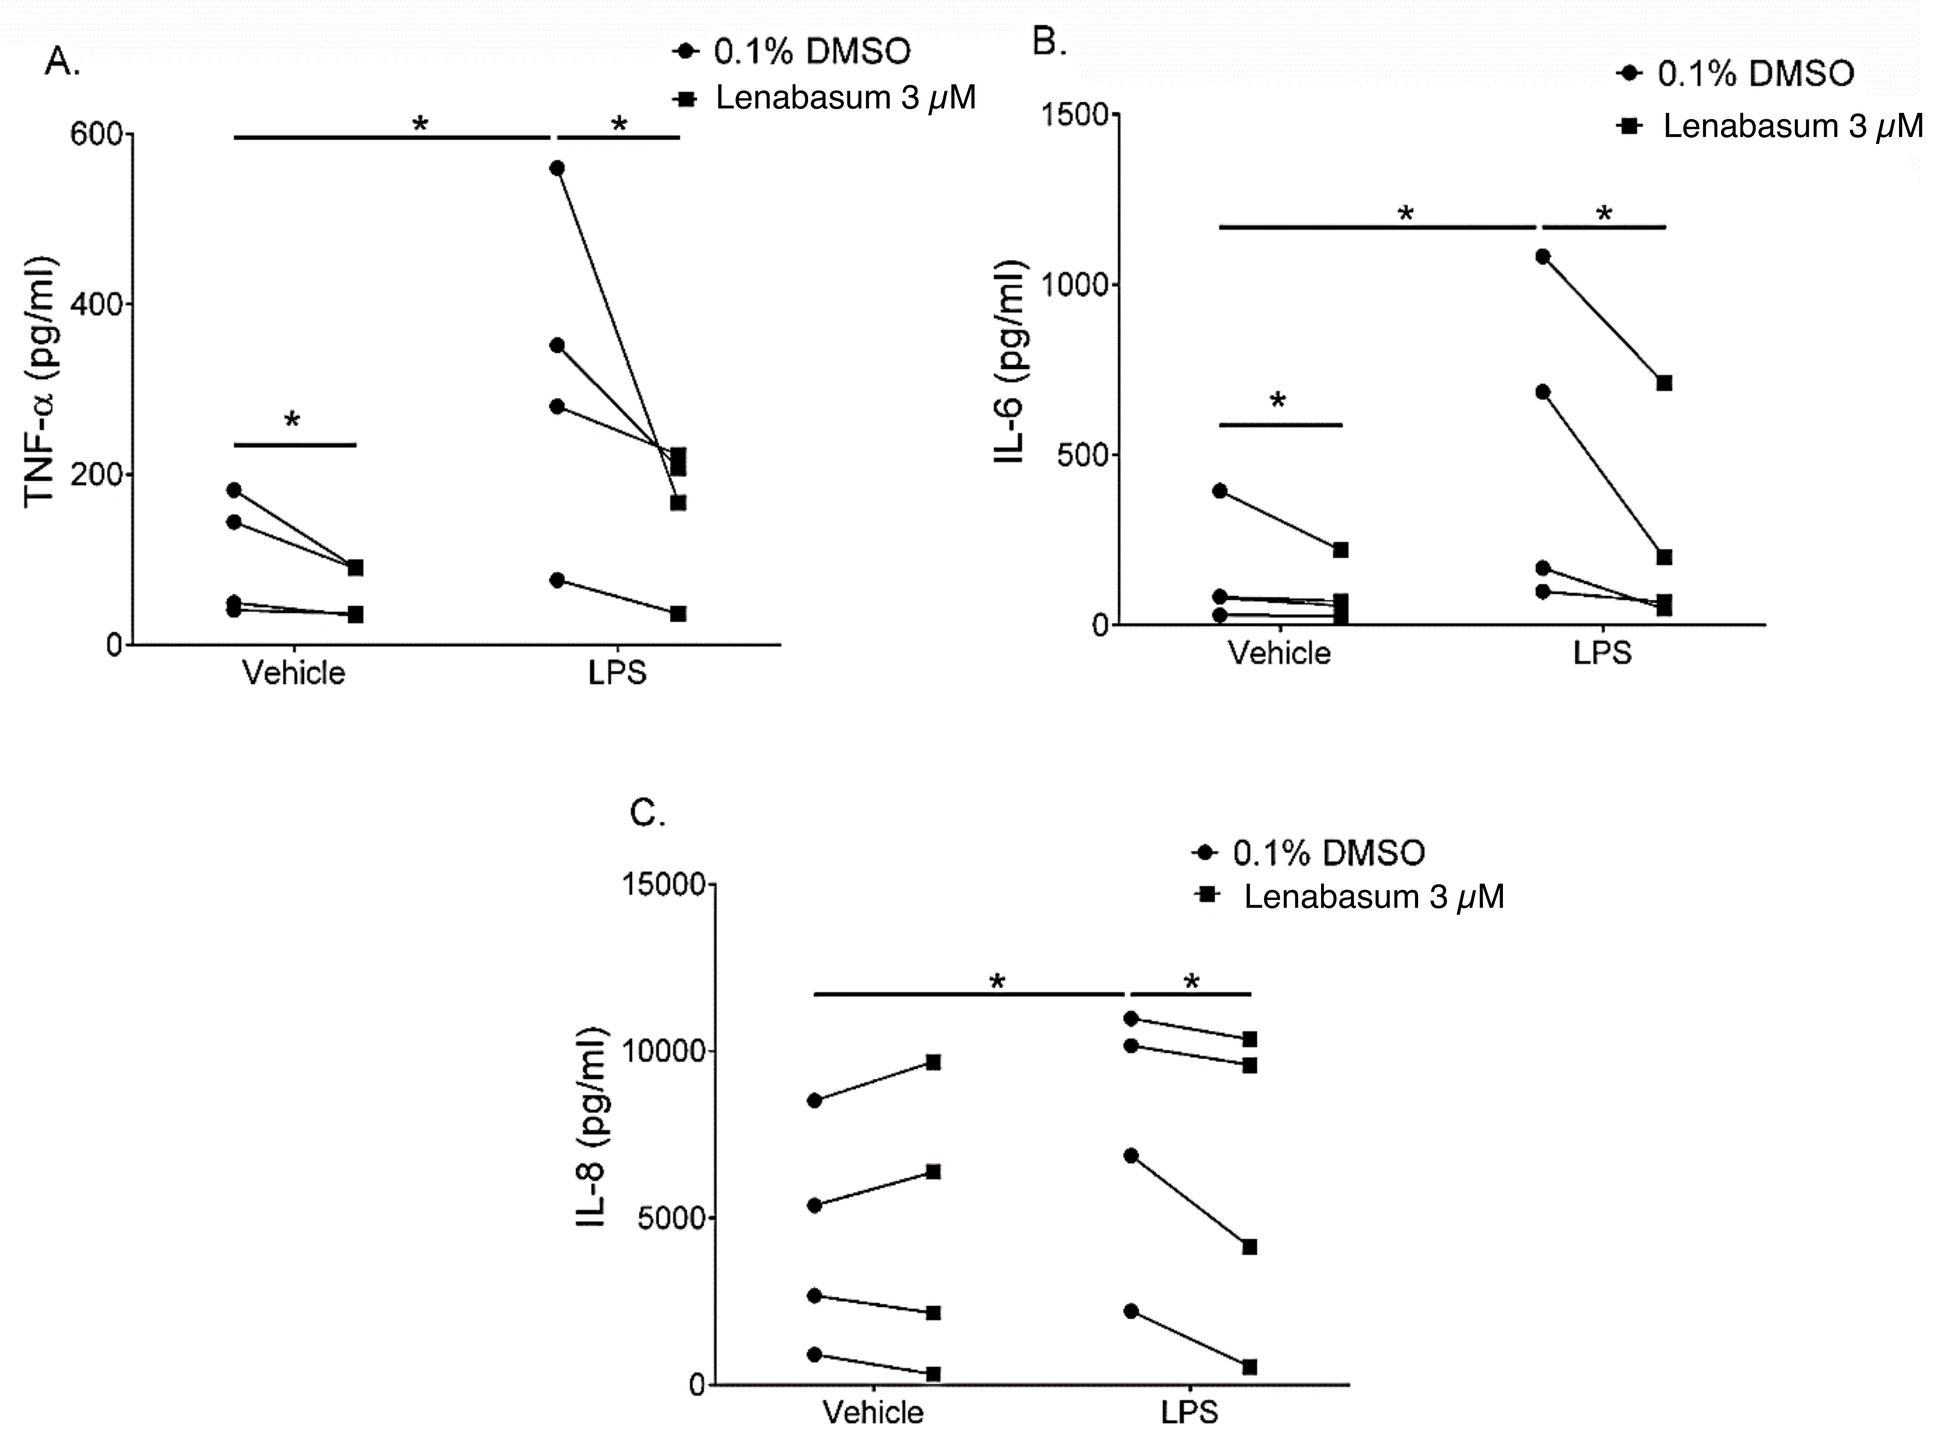
A.
0.1% DMSO
Lenabasum 3 μM
TNF-α (pg/ml)
Vehicle
LPS
B.
IL-6 (pg/ml)
C.
IL-8 (pg/ml)

Figure 1. Lenabasum decreases LPS-induced secretion of inflammatory mediators in primary cultures of CF AMs.
CF AMs were treated with 3 μM of Lenabasum for 6 h in presence of 0.1% DMSO (vehicle) or 100 ng/ml LPS from *P. aeruginosa*. TNF-α (A), IL-6 (B), and IL-8 (C) protein secretion into the culture media were determined by ELISA. Data are from 4 different samples before and after Lenabasum treatment in absence or presence of LPS. * p<0.05.

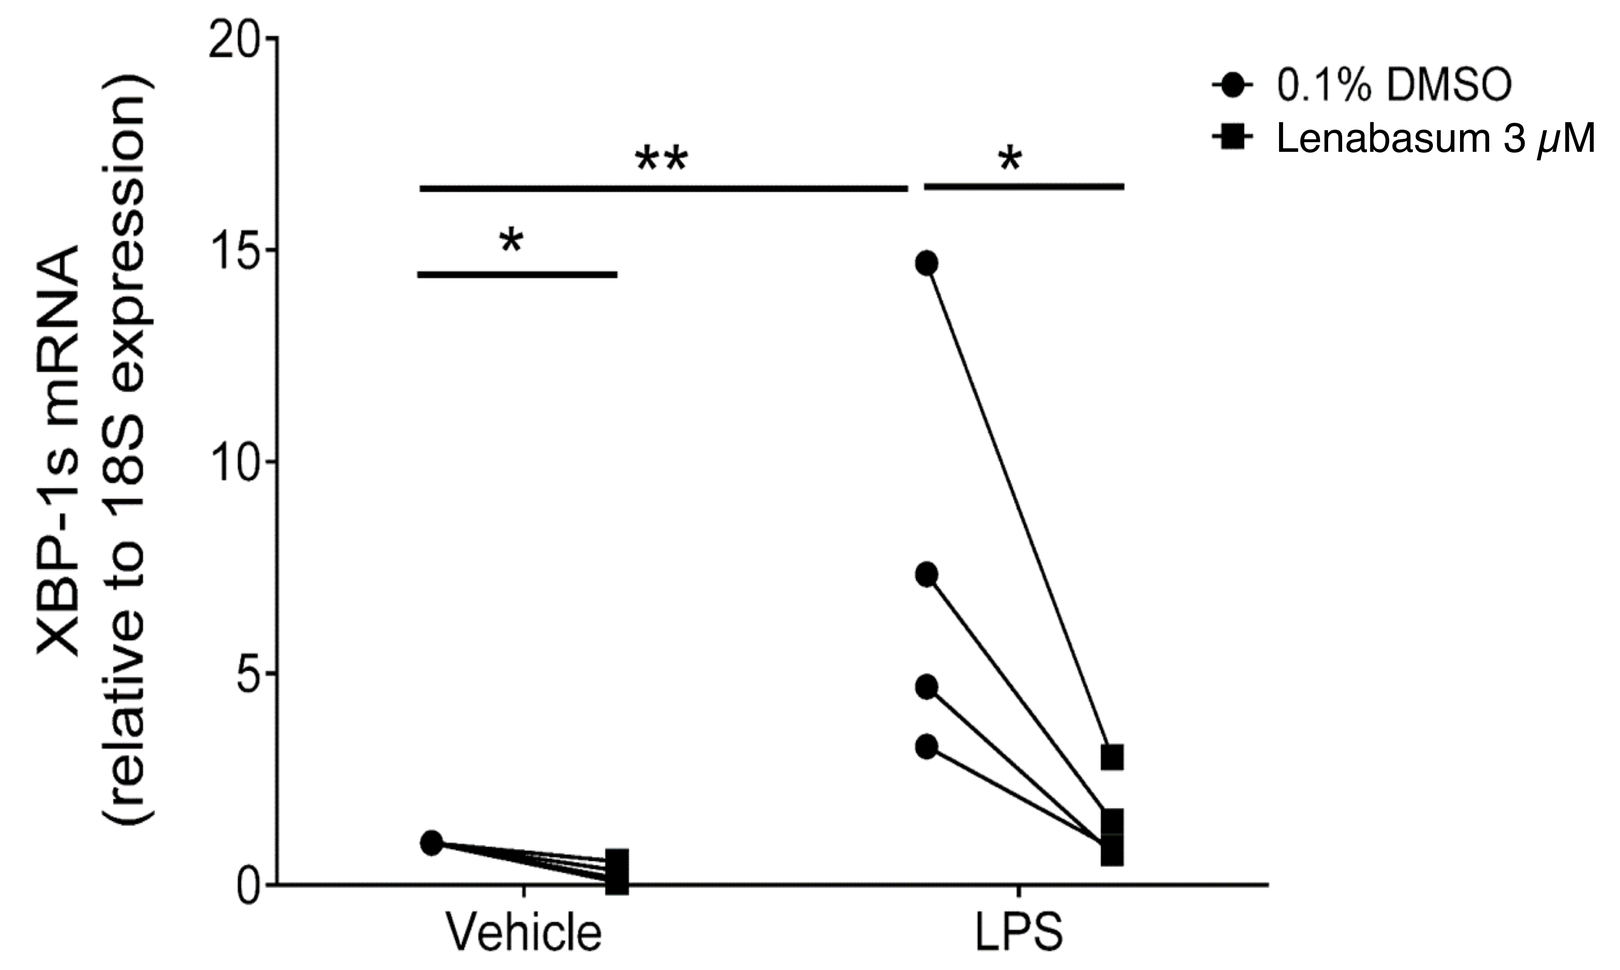
0.1% DMSO
Lenabasum 3 μM
XBP-1s mRNA
(relative to 18S expression)
Vehicle
LPS

Figure 2. Lenabasum decreases LPS-up-regulated XBP-1s levels in primary cultures of CF AMs.
CF AMs were treated with 3 μM of Lenabasum for 6 h in presence of 0.1% DMSO (vehicle) or 100 ng/ml LPS from *P. aeruginosa*. Quantitative RT-PCR was used to determine the mRNA levels of XBP-1s. Data are expressed as fold change relative to 18S mRNA. Data are from 4 different samples before and after Lenabasum treatment in absence or presence of LPS. * p<0.05 and ** p<0.01.

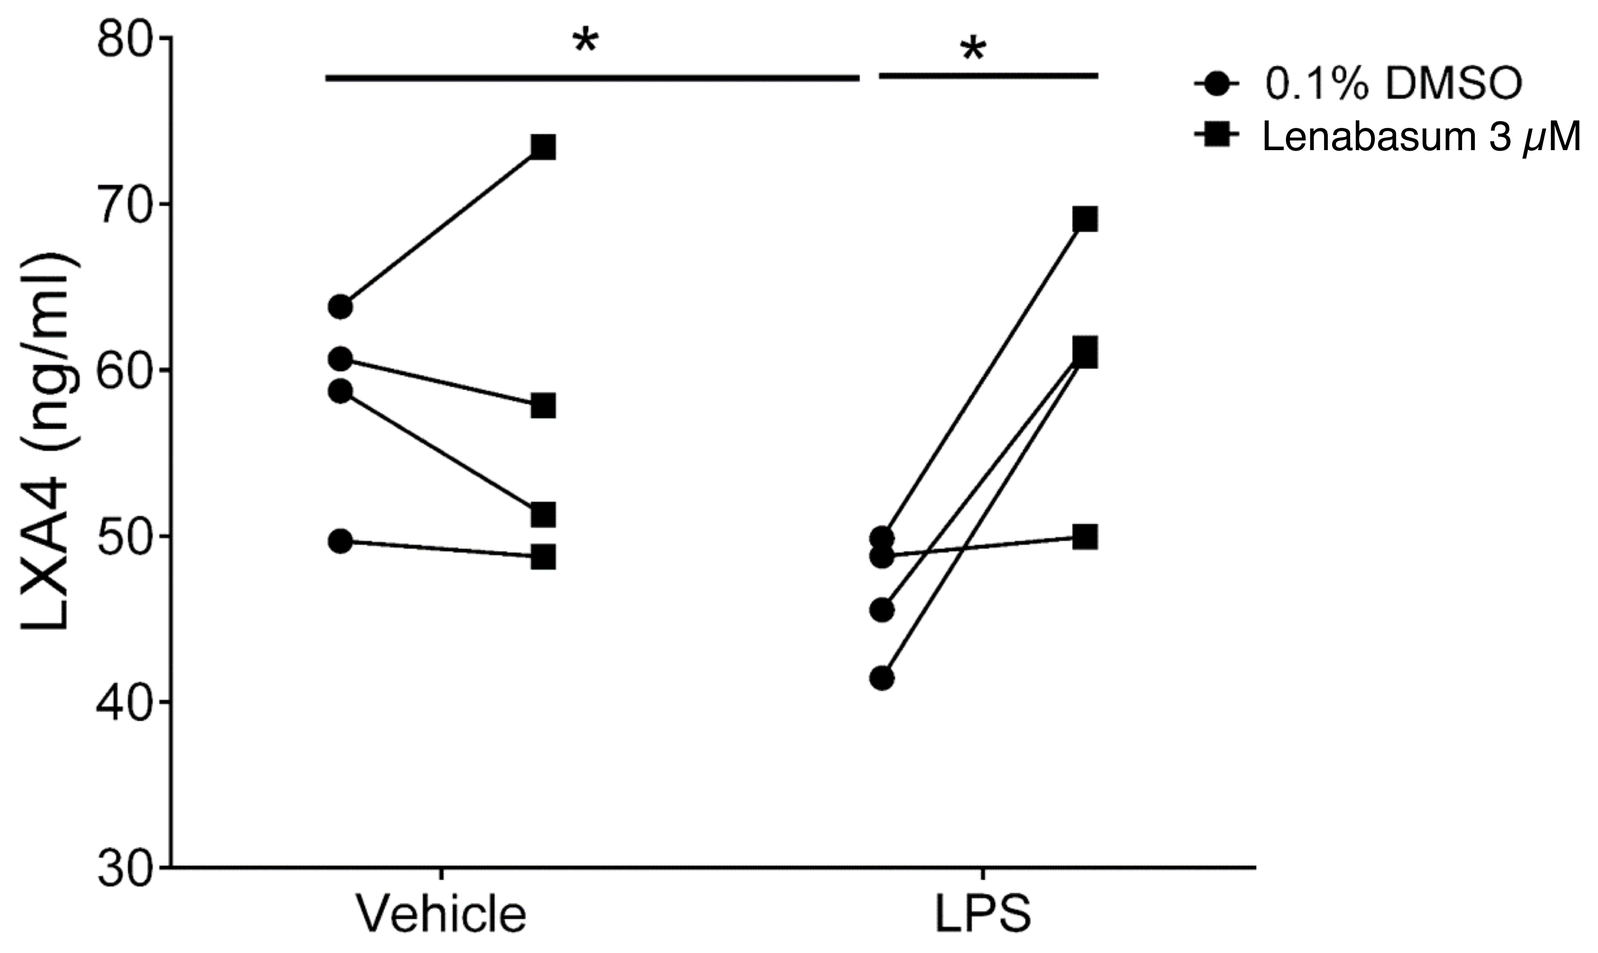
0.1% DMSO
Lenabasum 3 μM
LXA4 (ng/ml)
Vehicle
LPS

Figure 3. Lenabasum abrogates LPS-induced decreases in LXA4 in primary cultures of CF AMs.
CF AMs were treated with 3 μM of Lenabasum for 6 h in presence of 0.1% DMSO (vehicle) or 100 ng/ml LPS from *P. aeruginosa*. LXA4 secretion into the culture media was determined by ELISA. Data are from 4 different samples before and after Lenabasum treatment in absence or presence of LPS. * p<0.05.

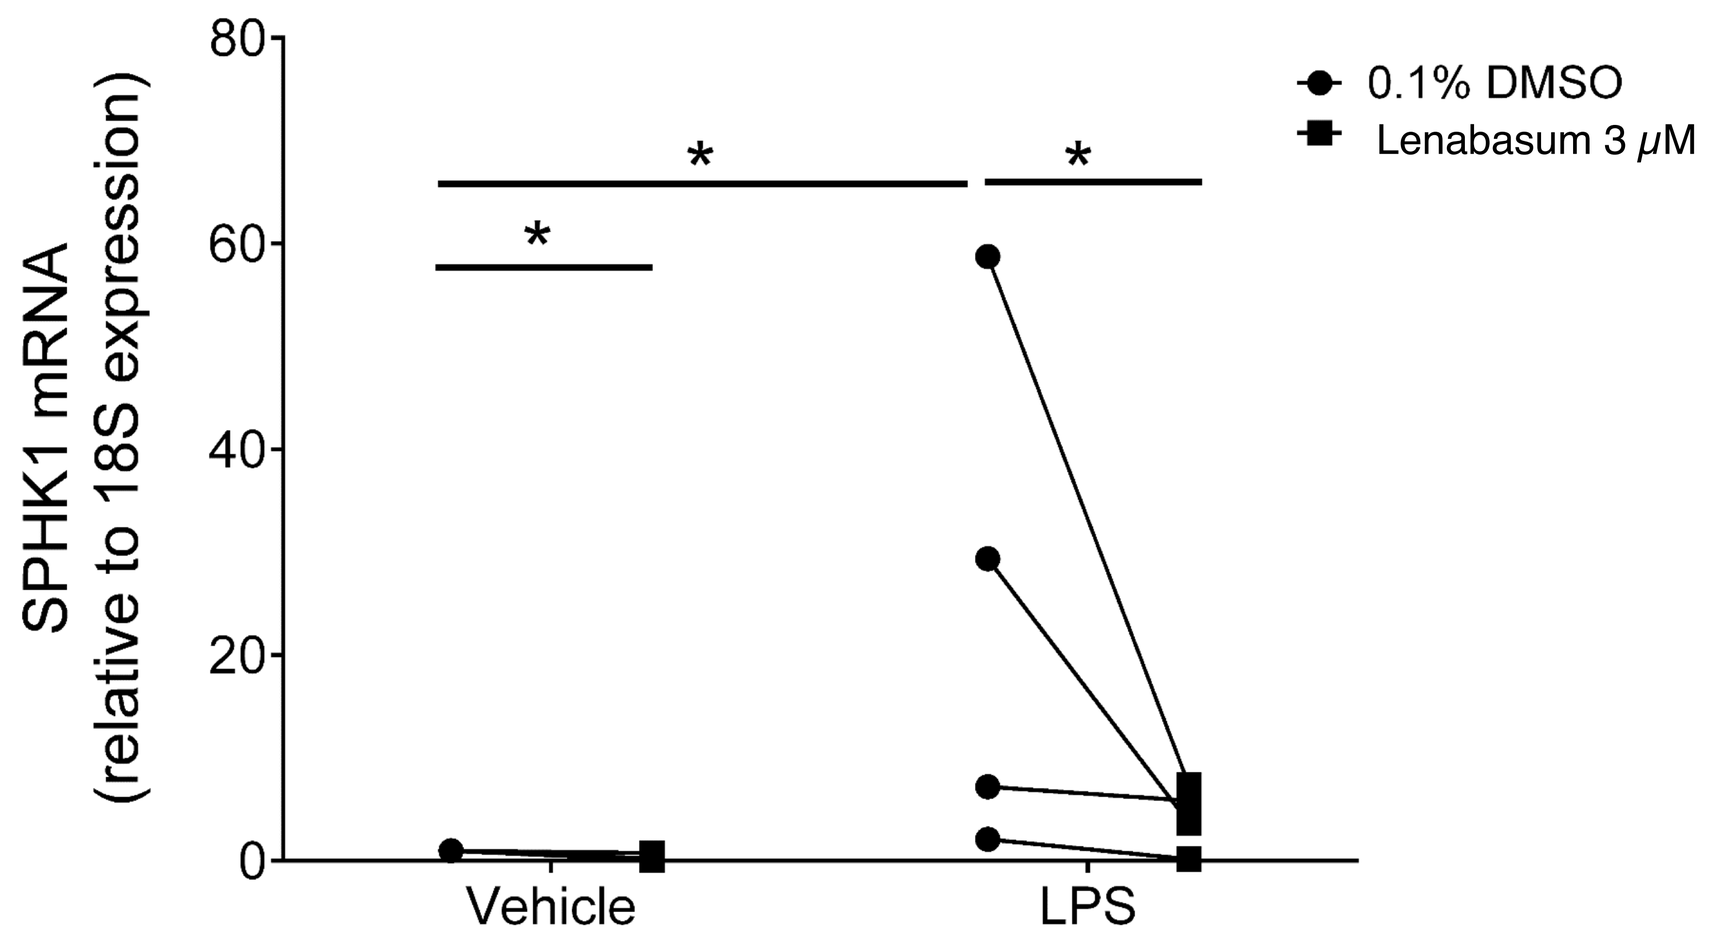
0.1% DMSO
Lenabasum 3 μM
SPHK1 mRNA
(relative to 18S expression)
Vehicle
LPS

Figure 4. Lenabasum decreases LPS-up-regulated sphingosine kinase 1 (SPHK1) in primary cultures of CF AMs.
CF AMs were treated with 3 μM of Lenabasum for 6 h in presence of 0.1% DMSO (vehicle) or 100 ng/ml LPS from *P. aeruginosa*. Quantitative RT-PCR was used to determine the mRNA levels of SPHK1. Data are expressed as fold change relative to 18S mRNA. Data are from 4 different samples before and after Lenabasum treatment in absence or presence of LPS. * p<0.05.

Results

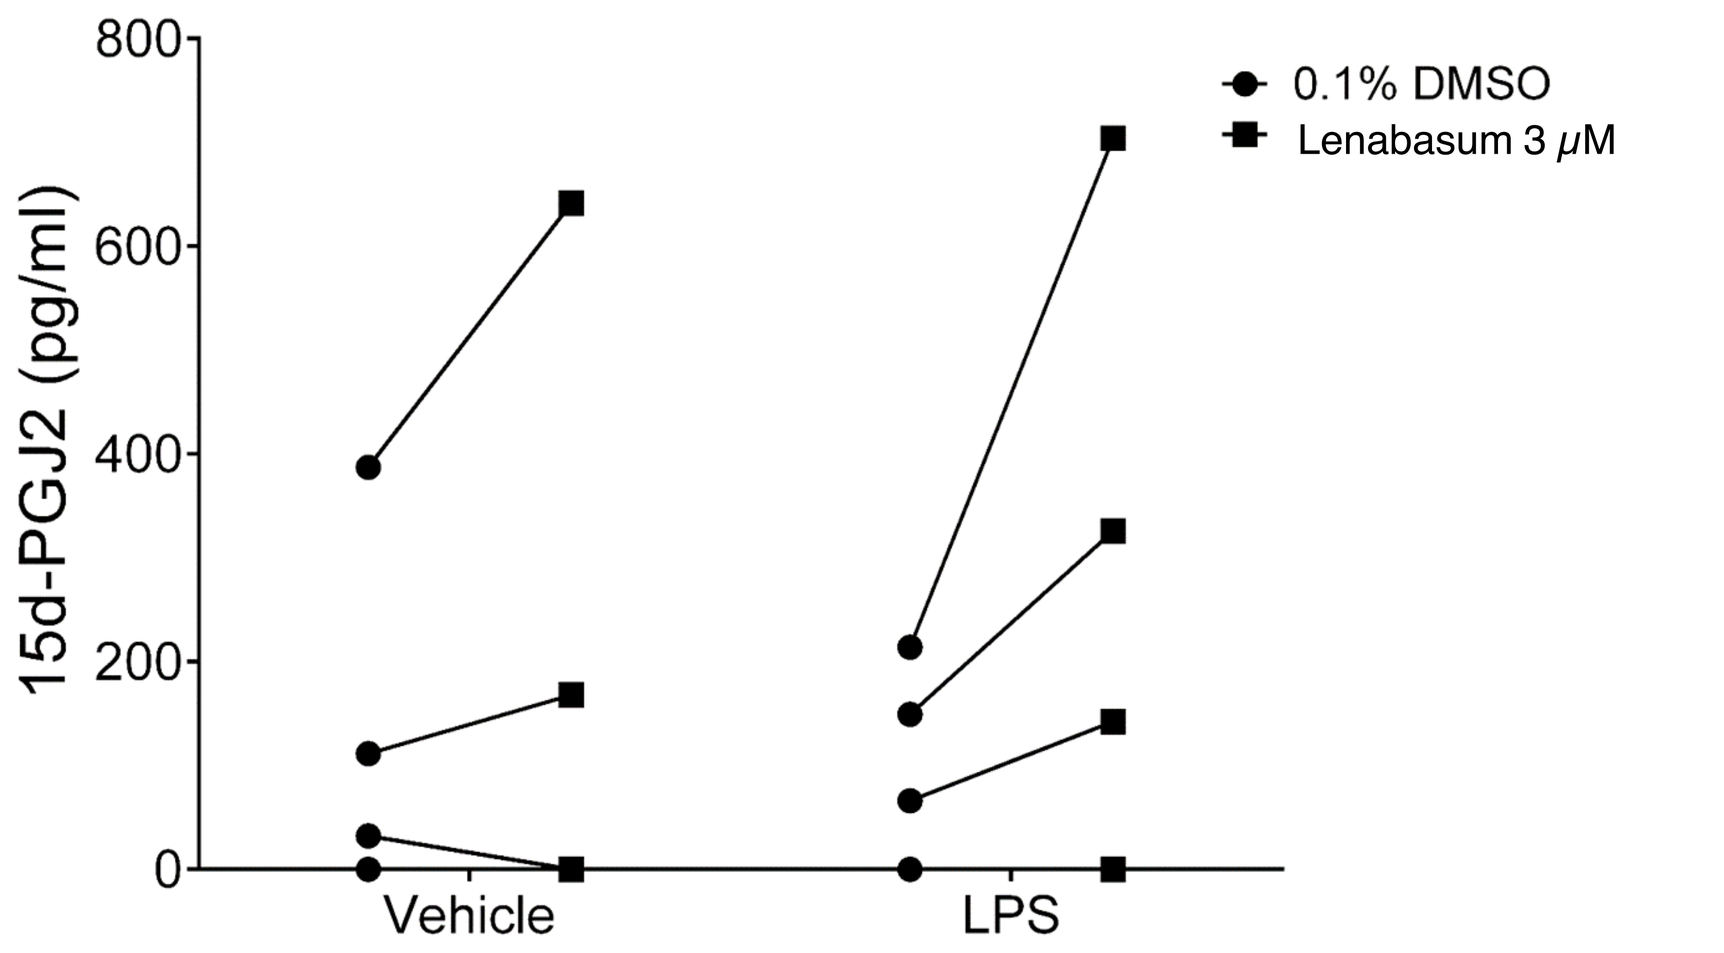
0.1% DMSO
Lenabasum 3 μM
15d-PGJ2 (pg/ml)
Vehicle
LPS

Figure 5. Lenabasum increases the levels of 15-deoxy-$\Delta^{12,14}$-PGJ2 in presence of LPS in primary cultures of CF AMs.
CF AMs were treated with 3 μM of Lenabasum for 6 h in presence of 0.1% DMSO (vehicle) or 100 ng/ml LPS from *P. aeruginosa*. 15d-PGJ2 secretion into the culture media were determined by ELISA. Data are from 4 different samples before and after Lenabasum treatment in absence or presence of LPS.

Biomarker	Baseline Values	LPS (100 ng/ml)			
			Lenabasum		
		0 µM	1 µM	3 µM	10 µM
XBP-1s mRNA (Relative to 18S expression)	1 ± 0.1	8.5 ± 1.1 #	2.6 ± 0.8 *	1.7 ± 0.5 *	2.9 ± 0.6 *
SPHK-1 mRNA (Relative to 18S expression)	1 ± 0.1	25.2 ± 5.3 #	3.2 ± 0.6 *	5.2 ± 1.3 *	3.8 ± 0.8 *
TNF-α (pg/ml)	88.3 ± 16.6	383.6 ± 99.4 #	150.2 ± 32.6 *	116.4 ± 26.5 *	149.6 ± 35.3 *
IL-8 (pg/ml)	2923.3 ± 939.5	6142.4 ± 1171.1 #	5222.2 ± 1277	4892 ± 1192.4 *	5076.7 ± 1218.5
15d-PGJ2 (pg/ml)	373.7 ± 160.6	387.2 ± 151.3	342.2± 138.3	638.1 ± 214 *	1027.6 ± 258.5 *

Table: Effect of lenabasum on LPS-induced inflammatory biomarkers in AMs of CF patients. n = 4-6 CF lungs. # P < 0.05, LPS vs. baseline; * P < 0.05, lenabasum + LPS vs. LPS.

Table 1. Dose response of Lenabasum on key inflammatory mediators in CF airway macrophages. CF AMs were treated with the indicated concentrations of Lenabasum for 6 h in presence of 0.1% DMSO (baseline) or 100 ng/ml LPS from *P. aeruginosa* (0, 1, 3 and 10μM). Cellular extracts or culture media were isolated and tested for the indicated analytes. Data are from 4 different samples before and after Lenabasum treatment in absence or presence of LPS.

Conclusion

Utilizing a translational model consisting of exposure of primary cultures of CF human AMs to LPS, our study indicates that lenabasum:

- Decreases inflammatory cytokines induced by LPS in CF human AMs.
- Decreases the levels of spliced XBP-1, a key transcription factor implicated in excessive inflammatory responses of CF human AMs.
- Triggers the biosynthesis of LXA4, a key pro-resolution mediator, in LPS-stimulated CF human AMs.
- Decreases the expression of sphingosine kinase 1, the rate limiting enzyme for generation of sphingosine 1-phosphase, a key mediator of inflammation.
- Triggers the biosynthesis of the pro-resolution eicosanoid 15-deoxy-$\Delta^{12,14}$-PGJ2, a natural PPAR-gamma agonist which acts to resolve inflammation.

These findings further suggest the beneficial therapeutic effects of lenabusum for CF airway inflammatory responses.

Acknowledgements

We thank Dr. Michael Knowles for scientific advice and critical discussions regarding this project.

This project was funded by Corbus Pharmaceuticals.

References

1. Lubamba BA, Jones LC, O'Neal WK, Boucher RC1, and Ribeiro CMP. X-Box-Binding Protein 1 and Innate Immune Responses of Human Cystic Fibrosis Alveolar Macrophages. Am J Respir Crit Care Med., 192:1449-61. 2015.
2. Zurier RB, Sun Y-P, George KL, Stebulis JA, Rossetti RG, Skulas A, Judge E, Serhan CN. Ajulemic acid, a synthetic cannabinoid, increases formation of the endogenous proresolving and anti-inflammatory eicosanoid, lipoxin A4. The FASEB Journal. 2009;23(5):1503–1509. doi:10.1096/fj.08-118323
3. Tepper MA, Zurier RB, Burstein SH. Ultrapure ajulemic acid has improved CB2 selectivity with reduced CB1 activity. Bioorganic & Medicinal Chemistry. 2014;22(13):3245–3251. doi:10.1016/j.bmc.2014.04.062
4. Motwani MP, Bennett F, Norris PC, Maini AA, George MJ, Newson J, Henderson A, Hobbs AJ, Tepper M, White B, et al. Potent Anti-Inflammatory and Pro-Resolving Effects of Anabasum in a Human Model of Self-Resolving Acute Inflammation. Clinical Pharmacology & Therapeutics. 2018;104(4):675–686. doi:10.1002/cpt.980