Resistance to APTO-253 caused by internal deletion and alternate promoter usage of the MYC gene in Raji B cells.

UC San Diego Health
MOORES CANCER CENTER

APTOSE BIOSCIENCES

#2096

Cheng-Yu Tsai[a], Andrea Local[b], Hongying Zhang[b], Stephen B. Howell[a,c] and William G. Rice[b]
[a]Moores Cancer Center, [b]Aptose Biosciences, Inc., [c]Department of Medicine, University of California, San Diego

INTRODUCTION

- **APTO-253 is a novel small molecule that inhibits expression of the MYC oncogene, leading to DNA damage, cell cycle arrest and apoptosis in human-derived solid tumor and hematologic cancer cells[1,2].**

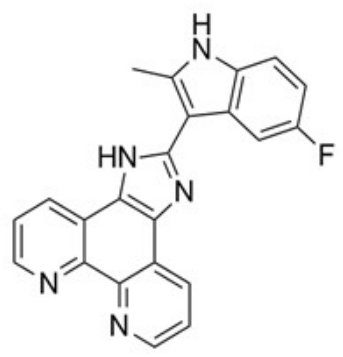

APTO-253

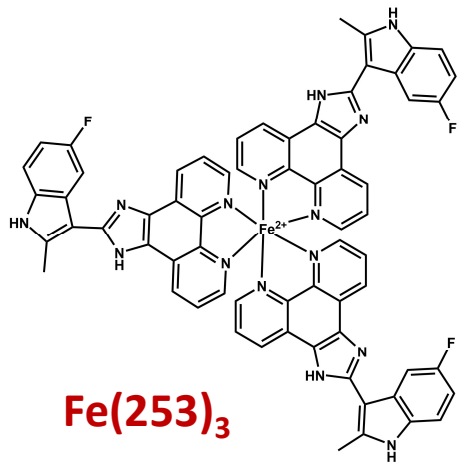

$Fe(253)_3$

- **In a Phase 1 trial in patients with solid tumors, APTO-253 was well tolerated and produced evidence of antitumor activity but did not cause myelosuppression even at the maximum tested dose.**
- **A Phase 1a/b trial of APTO-253 in patients with relapsed/refractory high risk MDS and AML is currently underway (NCT02267863).**
- **The purpose of this project was to understand how APTO-253 regulates MYC and how MYC regulation escapes as resistance emerges in B-cell lines.**

MATERIALS and METHODS

- **Cytotoxicity study** : Cell viability of primary patient cells and cultured cell lines was measured by MTS assay.
- **Selection of Raji cells for resistance**: The APTO-253-resistant Raji (Raji/253R) cell line was generated by exposure to progressively higher concentrations of APTO-253 over a period of 6 months.
- **RT-qPCR and RNA-seq**: Total cellular RNA was isolated using the RNeasy mini kit and cDNA was synthesized utilizing Transcriptor Universal cDNA master mix. Expression was calculated as fold change over control samples after normalizing to GAPDH (2^ΔΔCT). RNA-seq was performed at the UCSD IGM Genomics Center on an Illumina Sequencer HiSeq4000. sequence data was deposited to https://www.ncbi.nlm.nih.gov/geo/query/acc.cgi?acc=GSE111928
- **FRET assay:** FRET assay was performed by using dual labeled (5'FAM – 3'BHQ1) single stranded oligos. Melting temperature of each oligo was accessed in the presence of DMSO or escalating concentrations of APTO-253 using a Roche LightCycler 96 with 300s total incubation time at each temperature.
- **Flow cytometry for apoptosis cells:** Cells were stained with FITC-annexin V and propidium iodide and analyzed on BD Accuri C6 flow cytometer.
- **Statistical Analysis**: All two-group comparisons utilized Student's *t*-test with the assumption of unequal variance. Data are presented as mean ± SEM of a minimum of 3 independent experiments.

REFERENCES

1. Tsai C-Y et al., APTO-253 is a new addition to the repertoire of drugs that can exploit DNA BRCA1/2 deficiency. Mol Cancer Ther. 17(6):1167-1176, 2018.
2. Local A et al., APTO-253 stabilizes G-quadruplex DNA, inhibits MYC expression and induces DNA damage in acute myeloid leukemia cells. Mol Cancer Ther. 17(6):1177-1186, 2018.

RESULTS

- **APTO-253 potently kills hematologic malignant cell lines and primary samples from AML and CLL patients**

A

Disease Type	Cell lines	IC_{50} (µM) Mean
AML	MOLM-13	0.15
	MV4-11	0.23
	EOL-1	0.30
	Nomo-1	0.31
	THP-1	0.34
	HL-60	0.46
	SKM-1	0.47
	KG-1	0.50
	HEL92.1.7	1.75
B-ALL	RS4;11	0.03
	MHH-CALL-4	0.15
T-ALL	Jurkat	0.50
FL	DOHH2	0.11
MCL	Jeko-1	0.06
	GRANTA-519	0.09
	Mino	0.25
Burkitt's	RAJI	0.09
	Daudi	0.30
	Ramos	0.35
GCB-DLBCL	BJAB	0.30
	RL	0.32
	SU-DHL-6	0.49
ABC-DLBCL	U2932	0.06
	OCI-LY3	0.26
	SU-DHL-2	0.63
HL	L1236	0.25

B

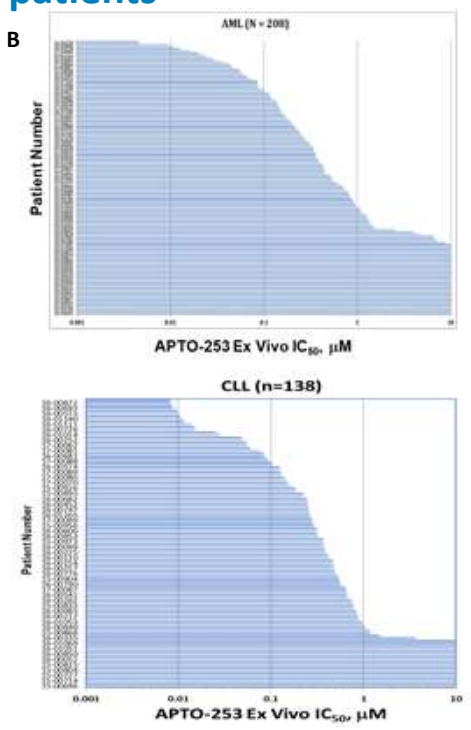

Patient Number
APTO-253 Ex Vivo IC_{50}, µM
CLL (n=138)
Patient Number
0.001
0.01
0.1
1
10
APTO-253 Ex Vivo IC_{50}, µM

(A) APTO-253 IC_{50} was measured in vitro against a panel of cancer cell lines by MTS assay (B) APTO-253 activity ex vivo against primary patients AML (top) and CLL (bottom) samples.

- **APTO-253 binds/stabilizes MYC DNA G-Quadruplex motif leading to inhibition of MYC gene expression and cell apoptosis**

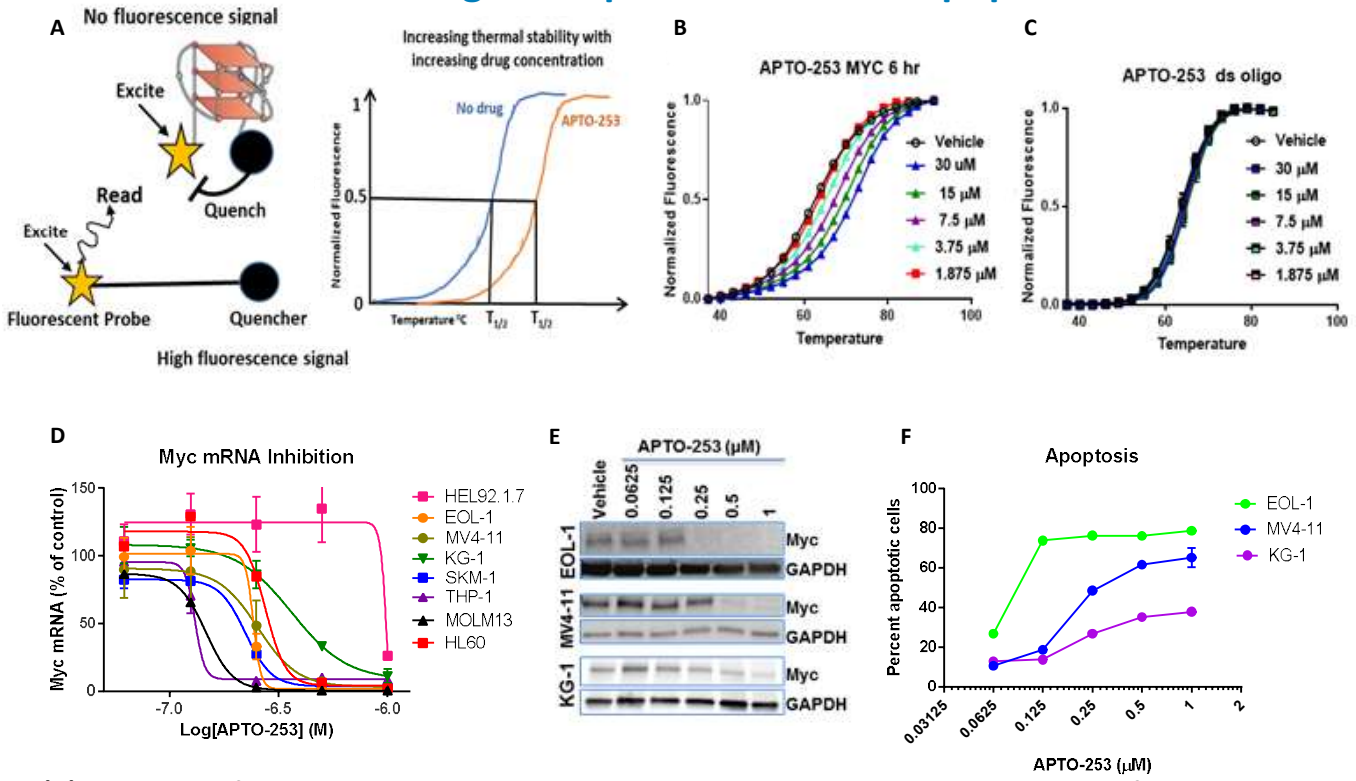

A
No fluorescence signal
Excite
Read
Quench
Excite
Fluorescent Probe
Quencher
High fluorescence signal
Increasing thermal stability with increasing drug concentration
No drug
APTO-253
Normalized Fluorescence
Temperature °C
B
APTO-253 MYC 6 hr
Normalized Fluorescence
Temperature
Vehicle
30 uM
15 µM
7.5 µM
3.75 µM
1.875 µM
C
APTO-253 ds oligo
Normalized Fluorescence
Temperature
Vehicle
30 µM
15 µM
7.5 µM
3.75 µM
1.875 µM
D
Myc mRNA Inhibition
Myc mRNA (% of control)
Log[APTO-253] (M)
HEL92.1.7
EOL-1
MV4-11
KG-1
SKM-1
THP-1
MOLM13
HL60
E
APTO-253 (µM)
Vehicle
0.0625
0.125
0.25
0.5
1
EOL-1
MV4-11
KG-1
Myc
GAPDH
F
Apoptosis
Percent apoptotic cells
APTO-253 (µM)
EOL-1
MV4-11
KG-1

(A) Schematic of quenching FRET assay. At low temperatures, the G-quadruplex structure forms and the fluorescent FAM signal is quenched by BHQ1; as the temperature is increased the G4 structure unfolds and the FAM signal increases. (B) Melting curves for MYC G4 oligos after 6 h incubation with APTO-253. (C) Melting curves of ds-DNA control oligos. (D) AML lines were treated for 24 h and MYC mRNA levels measured by RT-qPCR. (E) Western blot analysis of MYC protein level in EOL-1, MV4-11 and KG-1 cells treated for 24 h at the concentrations listed. (F) Percent of apoptotic EOL-1, MV4-11, and KG-1 cells after 24 h exposure to APTO-253.

- **Overexpression of ABCG2 renders Raji cells resistant to APTO-253**

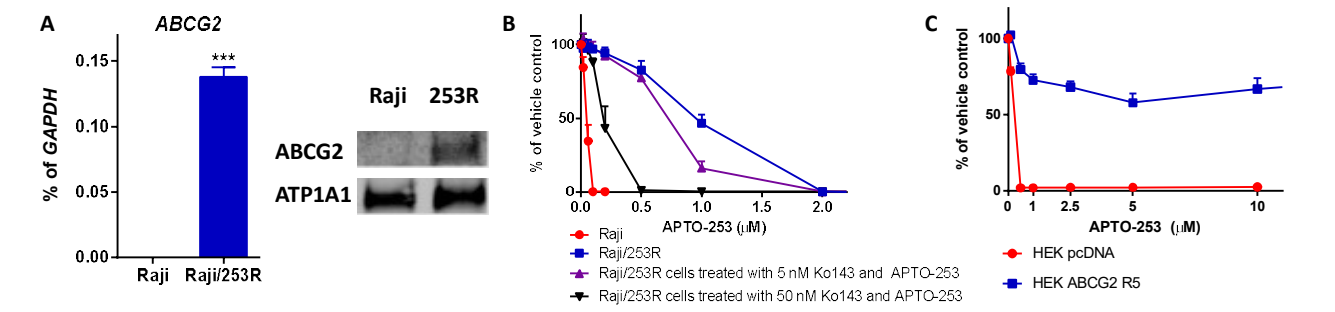

A
ABCG2
% of GAPDH

Raji
Raji/253R
Raji
253R
ABCG2
ATP1A1
B
% of vehicle control
APTO-253 (µM)
Raji
Raji/253R
Raji/253R cells treated with 5 nM Ko143 and APTO-253
Raji/253R cells treated with 50 nM Ko143 and APTO-253
C
% of vehicle control
APTO-253 (µM)
HEK pcDNA
HEK ABCG2 R5

(A) Relative levels of ABCG2 mRNA and protein in the Raji and Raji/253R. (B) Concentration-survival curves for Raji and Raji/253R treated with APTO-253 alone or in combination with APTO-253 and the ABCG2 inhibitor, Ko143 (C) Concentration-survival curves for HEK-293 transfected with pcDNA and ABCG2, clone R5 treated with APTO-253.

- **Resistance to APTO-253 caused by internal deletion and alternate promoter usage of the MYC gene**

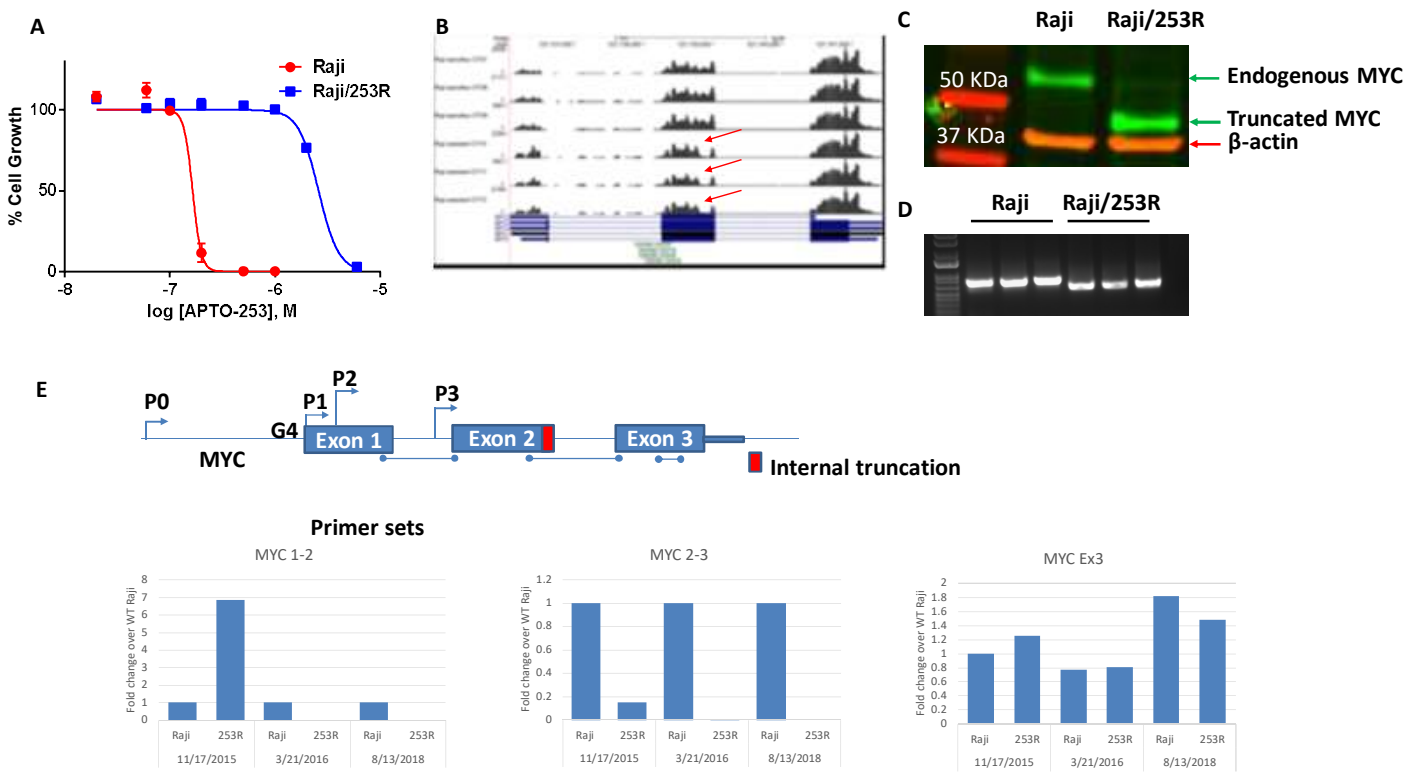

A
Raji
Raji/253R
% Cell Growth
log [APTO-253], M
B
C
Raji
Raji/253R
50 KDa
37 KDa
Endogenous MYC
Truncated MYC
β-actin
D
Raji
Raji/253R
E
P0
P1
P2
P3
G4
MYC
Exon 1
Exon 2
Exon 3
Internal truncation
Primer sets
MYC 1-2
MYC 2-3
MYC Ex3
Fold change over WT Raji
Raji
253R
11/17/2015
3/21/2016
8/13/2018

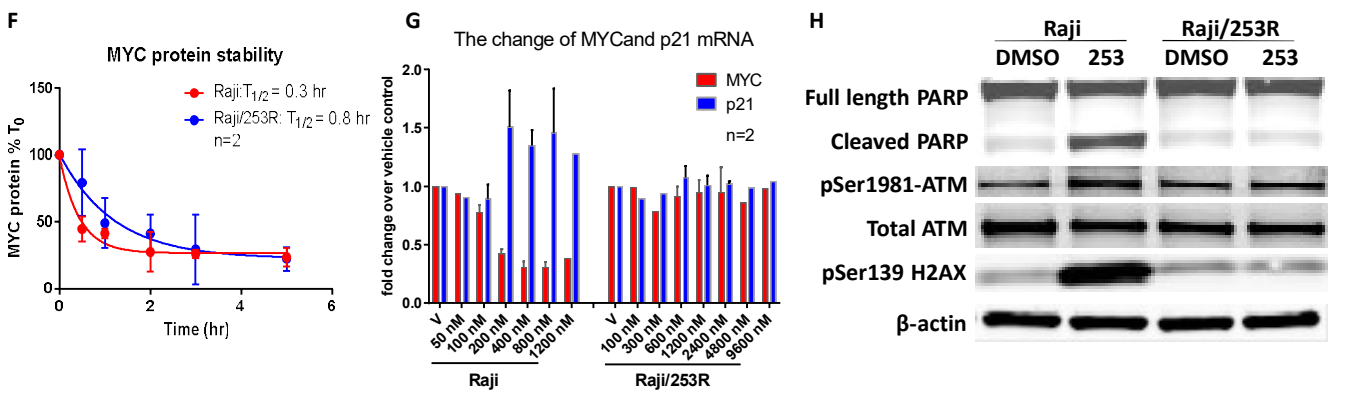

F
MYC protein stability
Raji: $T_{1/2}$ = 0.3 hr
Raji/253R: $T_{1/2}$ = 0.8 hr
n=2
MYC protein % T_0
Time (hr)
G
The change of MYCand p21 mRNA
MYC
p21
n=2
fold change over vehicle control
Raji
Raji/253R
H
Raji
Raji/253R
DMSO
253
DMSO
253
Full length PARP
Cleaved PARP
pSer1981-ATM
Total ATM
pSer139 H2AX
β-actin

(A) Concentration-survival curves for Raji and Raji/253R. (B) RNA-seq analysis revealed loss of a region of Exon 2. (C) Western blot analysis of MYC in the Raji and Raji/253R cells. (D) Genomic DNA isolated from the APTO-253 sensitive and resistant Raji cells was amplified by PCR with primers across Exon 2-Intron 2 boundary and the PCR products were run onto 1% agarose gel. (E) TOP: Primer sets design for MYC mRNA measurement. Bottom: MYC expression overtime with samples from 2015, 2016 and 2018 using different primer sets. (F) MYC protein stability in the Raji and Raji/253R cells. (G) Change of MYC and p21 transcripts in the Raji and Raji/253R cells treated with APTO-253. (H) Western blot analysis of proteins involved in apoptosis and DNA damage in Raji and Raji/253R treated with DMSO or APTO-253 0.5 µM for 24 h.

CONCLUSIONS

- APTO-253 potently kills malignant cells in both cell lines and primary patient samples.
- APTO-253 targets G-quadruplex motif in the P1/P2 promoter region of MYC gene and inhibits MYC gene expression to induce apoptosis.
- MYC driven Raji cells become resistant to APTO-253 via multiple mechanisms:
 - Up-regulation of ABCG2
 - Acquisition of a more stable MYC protein lacking the conserved core sequence of MYC Box III generated by deletion of internal region of MYC gene exon 2.
 - Utilization of an alternate P3 promoter not inhibited by G4 binding and stabilization
- Cells required three years and multiple modifications in MYC gene to generate high level drug resistance.
- Confirms essential role of MYC in the mechanism of APTO-253
- APTO-253 may serve as a safe and effective non-myelosuppressive first-in-class c-Myc inhibitor for treatment of hematologic malignancies including AML and CLL.