Abstract# PF337

STOCKHOLM 23RD CONGRESS JUNE 14 - 17 | 2018 European Hematology Association

CG'806, A NON-COVALENT PAN-FLT3/PAN-BTK INHIBITOR, EXHIBITS UNIQUE BINDING TO WILD TYPE AND C481S MUTANT BTK AND GREATER POTENC Y THAN IBRUTINIB AGAINST MALIGNANT B CELLS

Hongying Zhang[1], Andrea Local[1], Khalid Benbatoul[1], Peter Folger[1], Susan Sheng[1], Stephen E Kurtz[2], Jeffrey W. Tyner[3], Stephen B. Howell[4], William G. Rice[1]

[1]Aptose Biosciences, Inc, San Diego, CA, [2]Knight Cancer Institute, Division of Hematology and Medical Oncology, Oregon Health & Science University, [3]Department of Cell, Developmental & Cancer Biology, Oregon Health & Science University, Knight Cancer Institute, Portland, OR, [4]Moores Cancer Center, Department of Medicine, University of California, San Diego, La Jolla, United States

APTOSE BIOSCIENCES

INTRODUCTION

Bruton's tyrosine kinase (BTK) is a validated drug target due to its role in B-cell malignancy development. Ibrutinib, an irreversible BTK inhibitor that covalently binds to cysteine residue 481 (C481) and is approved for chronic lymphocytic leukemia (CLL) and other B-cell malignancies, is limited by its adverse effects and resistance resulting from C481S or other mutations. A safe and potent inhibitor against all forms of BTK is needed for patients intolerant, refractory and resistant to ibrutinib. CG'806 is an oral small molecule pan-FLT3/pan-BTK inhibitor, designed to solve ibrutinib's shortcomings. It is in development for acute myeloid leukemia (AML) and B-cell malignancies.

OBJECTIVES

We compared CG'806 and ibrutinib with respect to BTK binding mode, kinase inhibition profiles and cytotoxic activity against cultured and patient-derived malignant B-cells.

METHODS

- Crystallography: CG'806 was co-crystallized with the kinase domain of wild type (WT) and C481S mutant forms of BTK.
- Biochemical kinase inhibition assay: CG'806 was tested at 1 µM for biochemical inhibition of 583 kinases, and IC50s were determined on the most sensitive kinases with ATP concentration at Km and 1mM.
- The Safety44 panel (DiscoverX) was screened to identify potential off-target activities (ref #2).
- Cytotoxicity assay: cultured malignant B-cell lines, BTK transfected Ba/F3 cells or freshly isolated mononuclear cells from patients were treated with CG'806 or ibrutinib at the indicated concentration for 72 hr and MTS assay at the end.
- Western blotting: Vehicle or CG'806 treated cell lysates were assayed by Western blotting for cell signaling pathways.
- NFkB activity: nuclear extract of vehicle or CG'806 treated cells was assayed by TransAM NFkB colorimetric kit from Active Motif.

REFERENCES

1. Neuman L. et al., Blood, 2016, 128:2032
2. Bowes J, et al., Nat Rev Drug Discov. 2012 Dec;11(12):909-22

CG'806 Binds BTK WT and C481S Mutant as an Atypical Type II Inhibitor

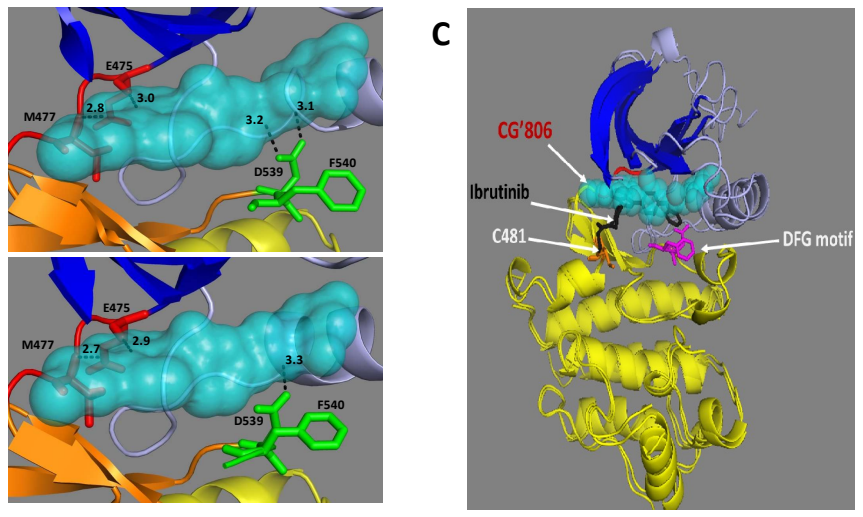
A
B
C
E475
M477
D539
F540
CG'806
Ibrutinib
C481
DFG motif

X-ray Crystal Structure of CG'806 in BTK WT and C481S mutant. The co-crystal complex of CG'806 bound to BTK- wild type (WT) or -C481S at resolution of 1.84Å and 1.63Å, respectively, revealed a binding mode of an atypical type II inhibitor. The DFG motif occupies an atypical conformation whereby the Phe540 is rotated out of the ATP binding pocket and the Asp539 side chain is tilted to hydrogen bond with CG'806. Moreover, CG'806 interacts with the hinge region and the αC-helix is in a partial out position. No electron density interaction was observed between CG'806 and the C481 residue. **A.** CG'806 binds BTK WT; **B.** CG'806 binds BTK C481S mutant; **C.** Overlay of CG'806 and ibrutinib binding in BTK WT.

CG'806 Inhibits BTK WT and C481S Mutants with Equal Potency

A

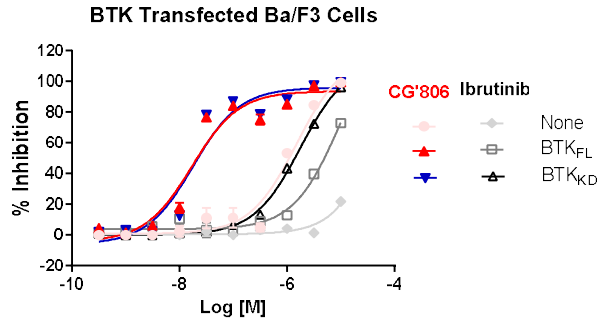
BTK Transfected Ba/F3 Cells
% Inhibition
Log [M]
CG'806
Ibrutinib
None
BTK FL
BTK KD

IC50 (nM)	CG'806	Ibrutinib
BTK_{FL}	22.8	4555.0
BTK_{KD}	23.2	1295.0
None	1001.0	> 10,000

B

Inhibitor	BTK binding profile	IC50 (nM), [ATP] = Km			IC50 (nM), [ATP] =1mM		
		BTK WT	BTK C481S	Fold (C481S/WT)	BTK WT	BTK C481S	Fold (C481S/WT)
CG806	non-covalent	5.0	2.5	0.5	9.3	13.1	1.4
Ibrutinib[1]	Covalent	0.1	6.6	66.0	NA	NA	NA

C

IC50 (nM)	Kinases related to Ibrutinib side effects			
	TEC	EGFR	ERBB2	ERBB4
CG806	>1,000	>1,000	>1,000	>1,000
Ibrutinib	78	5.6	9.4	NA

CG'806 potently inhibits BTK WT and C481S but not other kinases related to ibrutinib side effects.: A. CG'806 killed Ba/F3 cells transfected with BTK WT full length (FL) or kinase domain (KD) at equal potency, while ibrutinib had very weak killing effect; **B**. CG'806 has similar potency against BTK-WT and C481S mutant as opposed to ibrutinib that was >60-fold less potent against the C481S mutant. (Ibrutinib data are adapted from ref#1.) **. C.** CG'806 has no inhibitory effect on the kinases that related to ibrutinib side effects.

CG'806 Targets Specific Clusters of Oncogenic Kinases

A

Kinases	IC50 (nM), [ATP]=Km	IC50 (nM), [ATP]=1mM
Aurora A	0.4	25.5
FMS	0.6	44.5
TRKC	0.7	2.7
LCK	0.7	11.7
BLK	0.7	5.1
DDR2	1.0	8.1
MST1/STK4	2.1	6.7
Aurora C	2.9	14.8
Aurora B	3.7	32.7
ITK	4.3	14.4
TRKB	6.5	8.4
TIE2/TEK	6.5	4.6
LYN B	6.9	16.9
BTK	8.4	9.3
FLT3	8.7	8.1
c-MET	13.6	18.7
PDGFRa	14.0	25.8
BMX/ETK	14.5	9.1
TRKA	14.8	3.4
FER	14.8	33.5
RON/MST1R	34.5	25.6

B

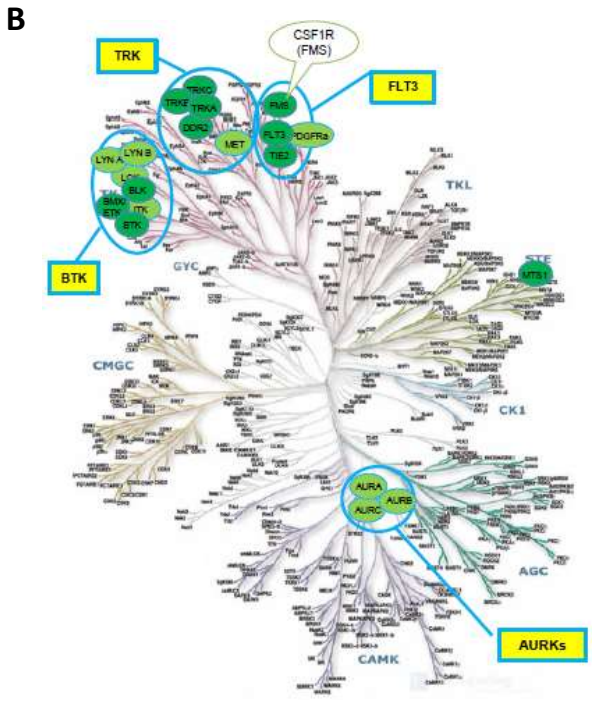
TRK
CSF1R (FMS)
FLT3
BTK
TKL
CMGC
CK1
AGC
CAMK
AURKs

Kinase profiling of CG'806: A. The kinases inhibited by CG'806 with IC50s < 50nM when biochemical enzymatic activity assays conducted at ATP=Km and 1mM; **B**. Kinome tree of CG'806 shows CG'806 most potently inhibits kinases from the BTK, FLT3, TRK, and AURK clusters with IC50s < 25 nM at 1mM ATP; **C.** CG'806 has similar potency against BTK-WT and C481S mutant as opposed to ibrutinib that was >60-fold less potent against the C481S mutant. (Ibrutinib data are adapted from ref#1.)

CG'806 Inhibits BCR, AKT/PI3K, NFkB Signaling in Malignant B-cells

CG'806 inhibited signaling from BCR, AKT/PI3K and NFkB and induced apoptosis in malignant B-cells: A. CG'806 decreased phosphorylation of BTK, ERK, AKT, PI3K and induced PARP cleavage in a cell line dependent manner. **B.** CG'806 decreased NFkB activity as represented by SU-DHL2 cells.

A

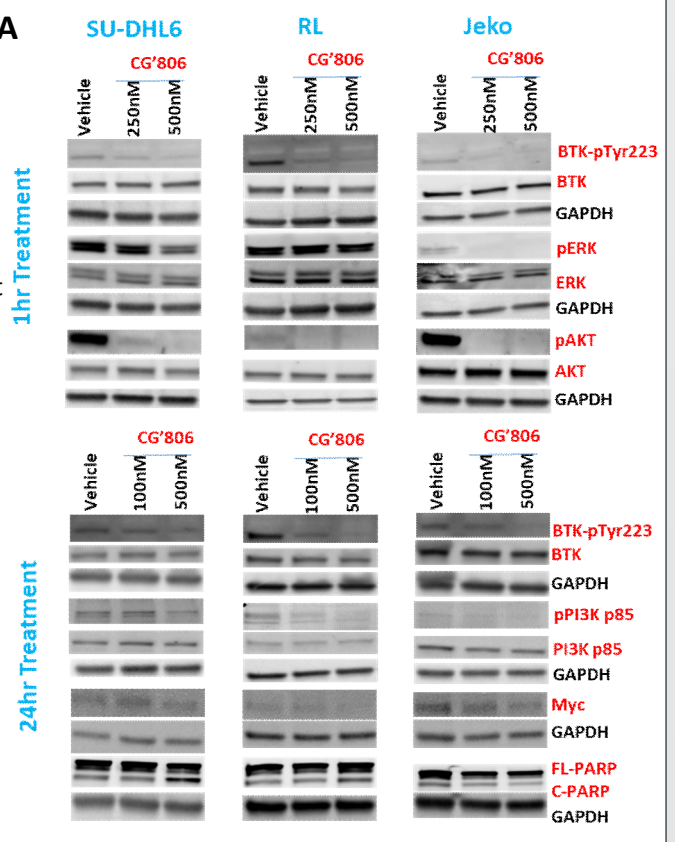
SU-DHL6
RL
Jeko
CG'806
Vehicle
250nM
500nM
100nM
1hr Treatment
24hr Treatment
BTK-pTyr223
BTK
GAPDH
pERK
ERK
pAKT
AKT
pPI3K p85
PI3K p85
Myc
FL-PARP
C-PARP

B

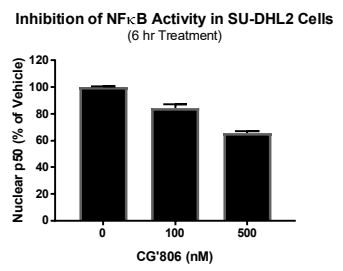
Inhibition of NFκB Activity in SU-DHL2 Cells
(6 hr Treatment)
Nuclear p50 (% of Vehicle)
CG'806 (nM)

CG'806 Kills Malignant B-cells More Effectively than Ibrutinib

A

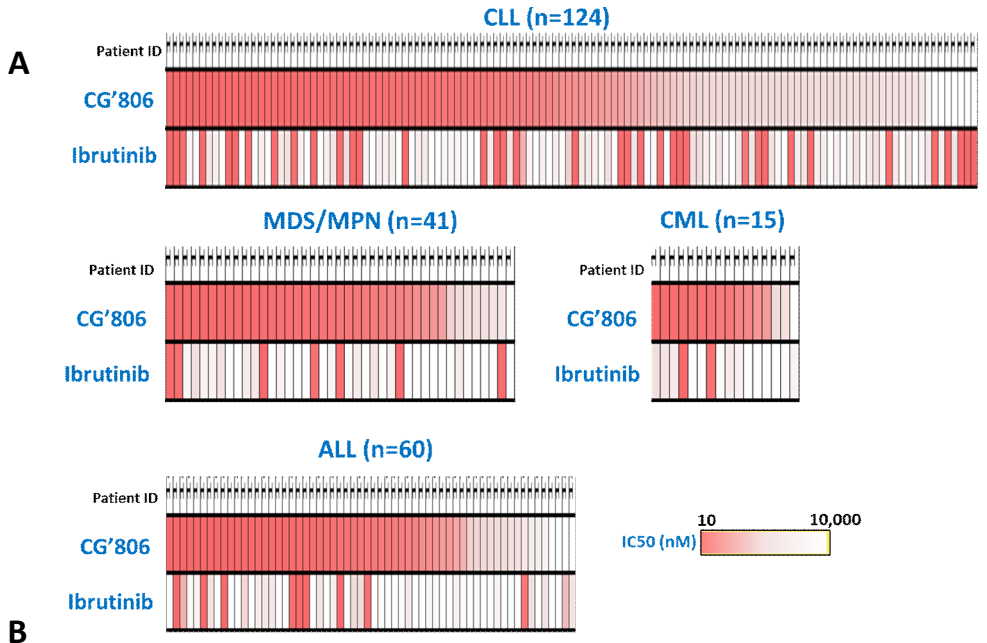
CLL (n=124)
MDS/MPN (n=41)
CML (n=15)
ALL (n=60)
Patient ID
CG'806
Ibrutinib
IC50 (nM)
10
10,000

B

Disease Type	Cell Line	IC_{50} (µM) CG'806	IC_{50} (µM) Ibrutinib	Fold difference
MCL	Mino	0.006	18.2	3033
	Granta-519	0.020	29.9	1495
	Jeko-1	0.197	23.6	120
B-ALL	RS411	0.002	12.4	6200
	MHH-Call4	0.026	22.8	877
Burkitt's	Ramos	0.014	22.0	1571
	Daudi	0.236	23.9	101
ABC-DLBCL	OCI-Ly3	0.830	29.0	35
	U2932	0.632	8.9	14
	SU-DHL2	0.744	27.0	36
GCB-DLBCL	BJAB	0.882	1.8	2
	SU-DHL6	0.083	1.2	14
	RL	0.667	17.0	25
FL	DOHH2	0.003	0.441	147

CG'806 has greater potency than ibrutinib: A. Primary CLL, ALL, CML, MDS/MPN samples were significantly more sensitive to CG'806 as compared to ibrutinib; **B.** CG'806 inhibited cell proliferation 2-6,000 times (Mean=976) more potently than ibrutinib in 14 tested malignant B-cell lines.

CONCLUSIONS

- **CG'806 is a potent, non-covalent, oral, atypical type II inhibitor of WT and C481S BTK.**
- **CG'806 inhibits BTK-C481S mutant and BTK-WT equivalently.**
- **CG'806 targets specific clusters of oncogenic kinases and inhibits signaling from BCR, AKT/PI3K and NFkB to induce apoptosis in malignant B cells.**
- **CG'806 killed cultured and primary malignant B-cells more potently than ibrutinib without affecting TEC, EGFR, ERBB2/4 or other safety-related targets.**
- **CG'806, as a potent pan-FLT3/pan-BTK inhibitor that is well tolerated in murine xenograft models, is suitable for development in patients with CLL and other B-cell malignancies intolerant, resistant, or refractory to ibrutinib.**