4626

KNIGHT CANCER INSTITUTE
Oregon Health & Science University

CG’806, a First-in-Class FLT3/BTK Inhibitor, Exhibits Potent Growth Inhibition as a Single Agent and in Combination with a BET bromodomain Inhibitor or a BCL2 Inhibitor on Primary AML and CLL Patient Samples

Stephen E. Kurtz, PhD[1], Beth Wilmot, PhD[2], Shannon K. McWeeney, PhD[2], Andrea Local, PhD[3], Hongying Zhang, MD, PhD[3], Stephen B. Howell, MD[3], William G. Rice, PhD[3], Brian J. Druker, MD[1,4], and Jeffrey W. Tyner, PhD[5]

[1] *Division of Hematology and Medical Oncology, Knight Cancer Institute, Oregon Health & Science University, Portland, Oregon;* [2] *Division of Bioinformatics and Computational Biology, Oregon Health & Science University, Portland, Oregon,* [3]Aptose Biosciences, San Diego, CA. [4] *Howard Hughes Medical Institute, Portland, Oregon;* [5] *Department of Cell, Developmental, and Cancer Biology, Oregon Health & Science University*

ABSTRACT

Background: One of the daunting challenges for the use of targeted therapies in the treatment of acute *Introduction*: Acute myeloid leukemia (AML) and chronic lymphocytic leukemia (CLL) are two of the most common adult hematologic malignancies. Cytogenetic and deep-sequencing studies of AML have revealed a complex and heterogeneous disease, with nearly 2,000 somatically mutated genes observed across the TCGA cohort of 200 patients, where nearly 200 of these somatic mutations were observed recurrently. Although the standard of care for AML has remained unchanged for decades, the development of FLT3 inhibitors represents a focused attempt at new therapies, as gain-of-function point mutation or internal tandem duplication (ITD) of FLT3 occur in ~30% of AML patients. Numerous FLT3 inhibitors have been developed with several showing transient clinical benefit, but responses have only lasted 3-4 months. Similarly, a majority of CLL patients develop resistance to treatment and remain incurable with current therapies. Recent insights into the importance of B-cell receptor-mediated kinase signaling pathways have resulted in new therapeutic strategies for CLL patients, including agents that target the kinases PI3K, SYK, and BTK. These agents have shown clinical activity, but responses have not been uniform, and the majority of patients eventually develop drug resistance. Thus, both AML and CLL are in urgent need of effective therapies that provide more durable clinical responses, which will most certainly involve novel drug combinations.

CG’806 is a first-in-class small molecule multi-kinase inhibitor against wild type (WT) and mutant forms of the FLT3 and Bruton’s tyrosine kinase (BTK) enzymes. We previously demonstrated that CG’806 has inhibitory activity against Ba/F3 cells expressing FLT3-ITD or point mutations, including D835G, D835Y, D835H, and F691L. We also evaluated the potency of CG’806 on various hematologic malignancy cell lines and patient primary bone marrow specimens. CG’806 exhibits broad and potent activity against primary patient samples over a range of hematologic malignancy subtypes, including AML, CLL, myelodysplastic syndromes/myeloproliferative neoplasms (MDS/MPN), and acute lymphoblastic leukemia (ALL). Preliminary analyses reveal a trend of greater sensitivity to CG’806 in FLT3-mutant AML cases compared with wild-type FLT3, although ongoing accrual of additional patient samples will be required to sufficiently power a statistical association of CG’806 sensitivity with FLT3 mutational status.

EX VIVO FUNCTIONAL ASSAY

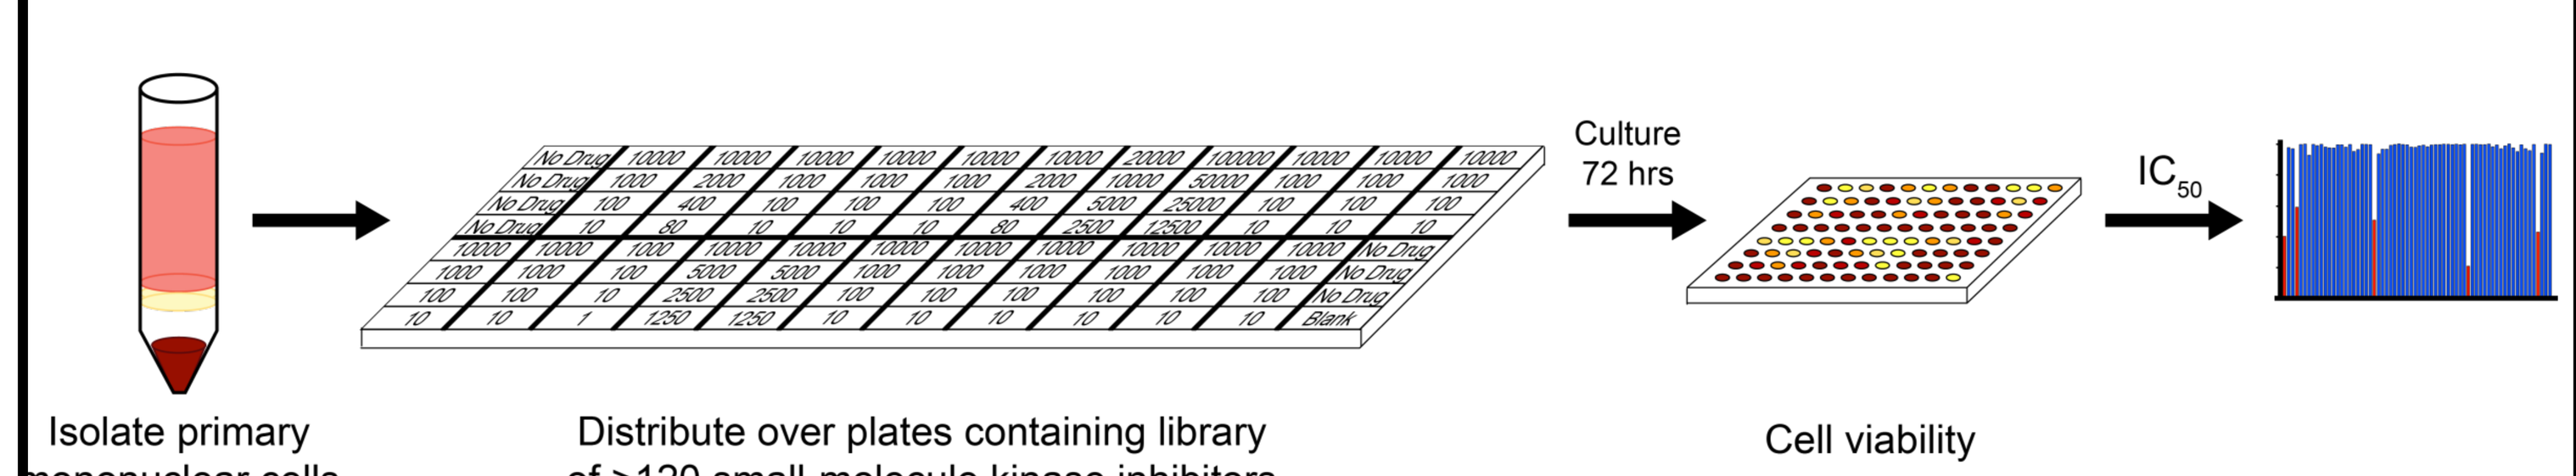
Isolate primary mononuclear cells
Distribute over plates containing library of >120 small-molecule kinase inhibitors
Culture 72 hrs
Cell viability assay
IC50

- 384-well format using 10,000 cells/well
- Cell viability (MTS) performed on day 3
- Combinations tested in fixed molar concentration series

***Ex Vivo Drug Sensitivity* Assay**

We used an *ex vivo* drug sensitivity assay to determine the activity of CG’806 as a single agent and in combination with the BET bromodomain inhibitor OTX-015 or the BCL2 inhibitor venetoclax on freshly isolated primary patient samples. Combinations were tested at fixed, equimolar ratios over the same dose range. After a 3-day *ex vivo* culture, cell viability was assessed using a colorimetric, tetrazolium-based MTS assay and IC_{50} values calculated. Sensitivity to CG’806 was evaluated across a range of concentrations after 72-hour treatment. IC_{50} was calculated as a measure of drug sensitivity.

__Disclosures:__ A.L. And H.Z are employees of Aptose Bioseceinces. SBH serves on the Advisory Board for Aptose Biosceiences. WGR is an employee of and has equity ownership in Aptose Biosceinces. J.W.T. receives research funding from Aptose Biosceinces. B.J.D. serves on the Advisory Board for Aptose Biosceinces and has equity ownership in Aptose Biosceinces. __Funding:__ Leukemia & Lymphoma Society

Sensitivity of CG’806 on Primary patient samples

DRUG	DIAGNOSIS SUBGROUPS (# Patient Samples Tested)			
	AML	CLL	MDS/MPN	ALL
CG'806	204	105	33	38
CG'806 + OTX-015	84	24	13	19
CG'806 + Venetoclax	84	24	13	19

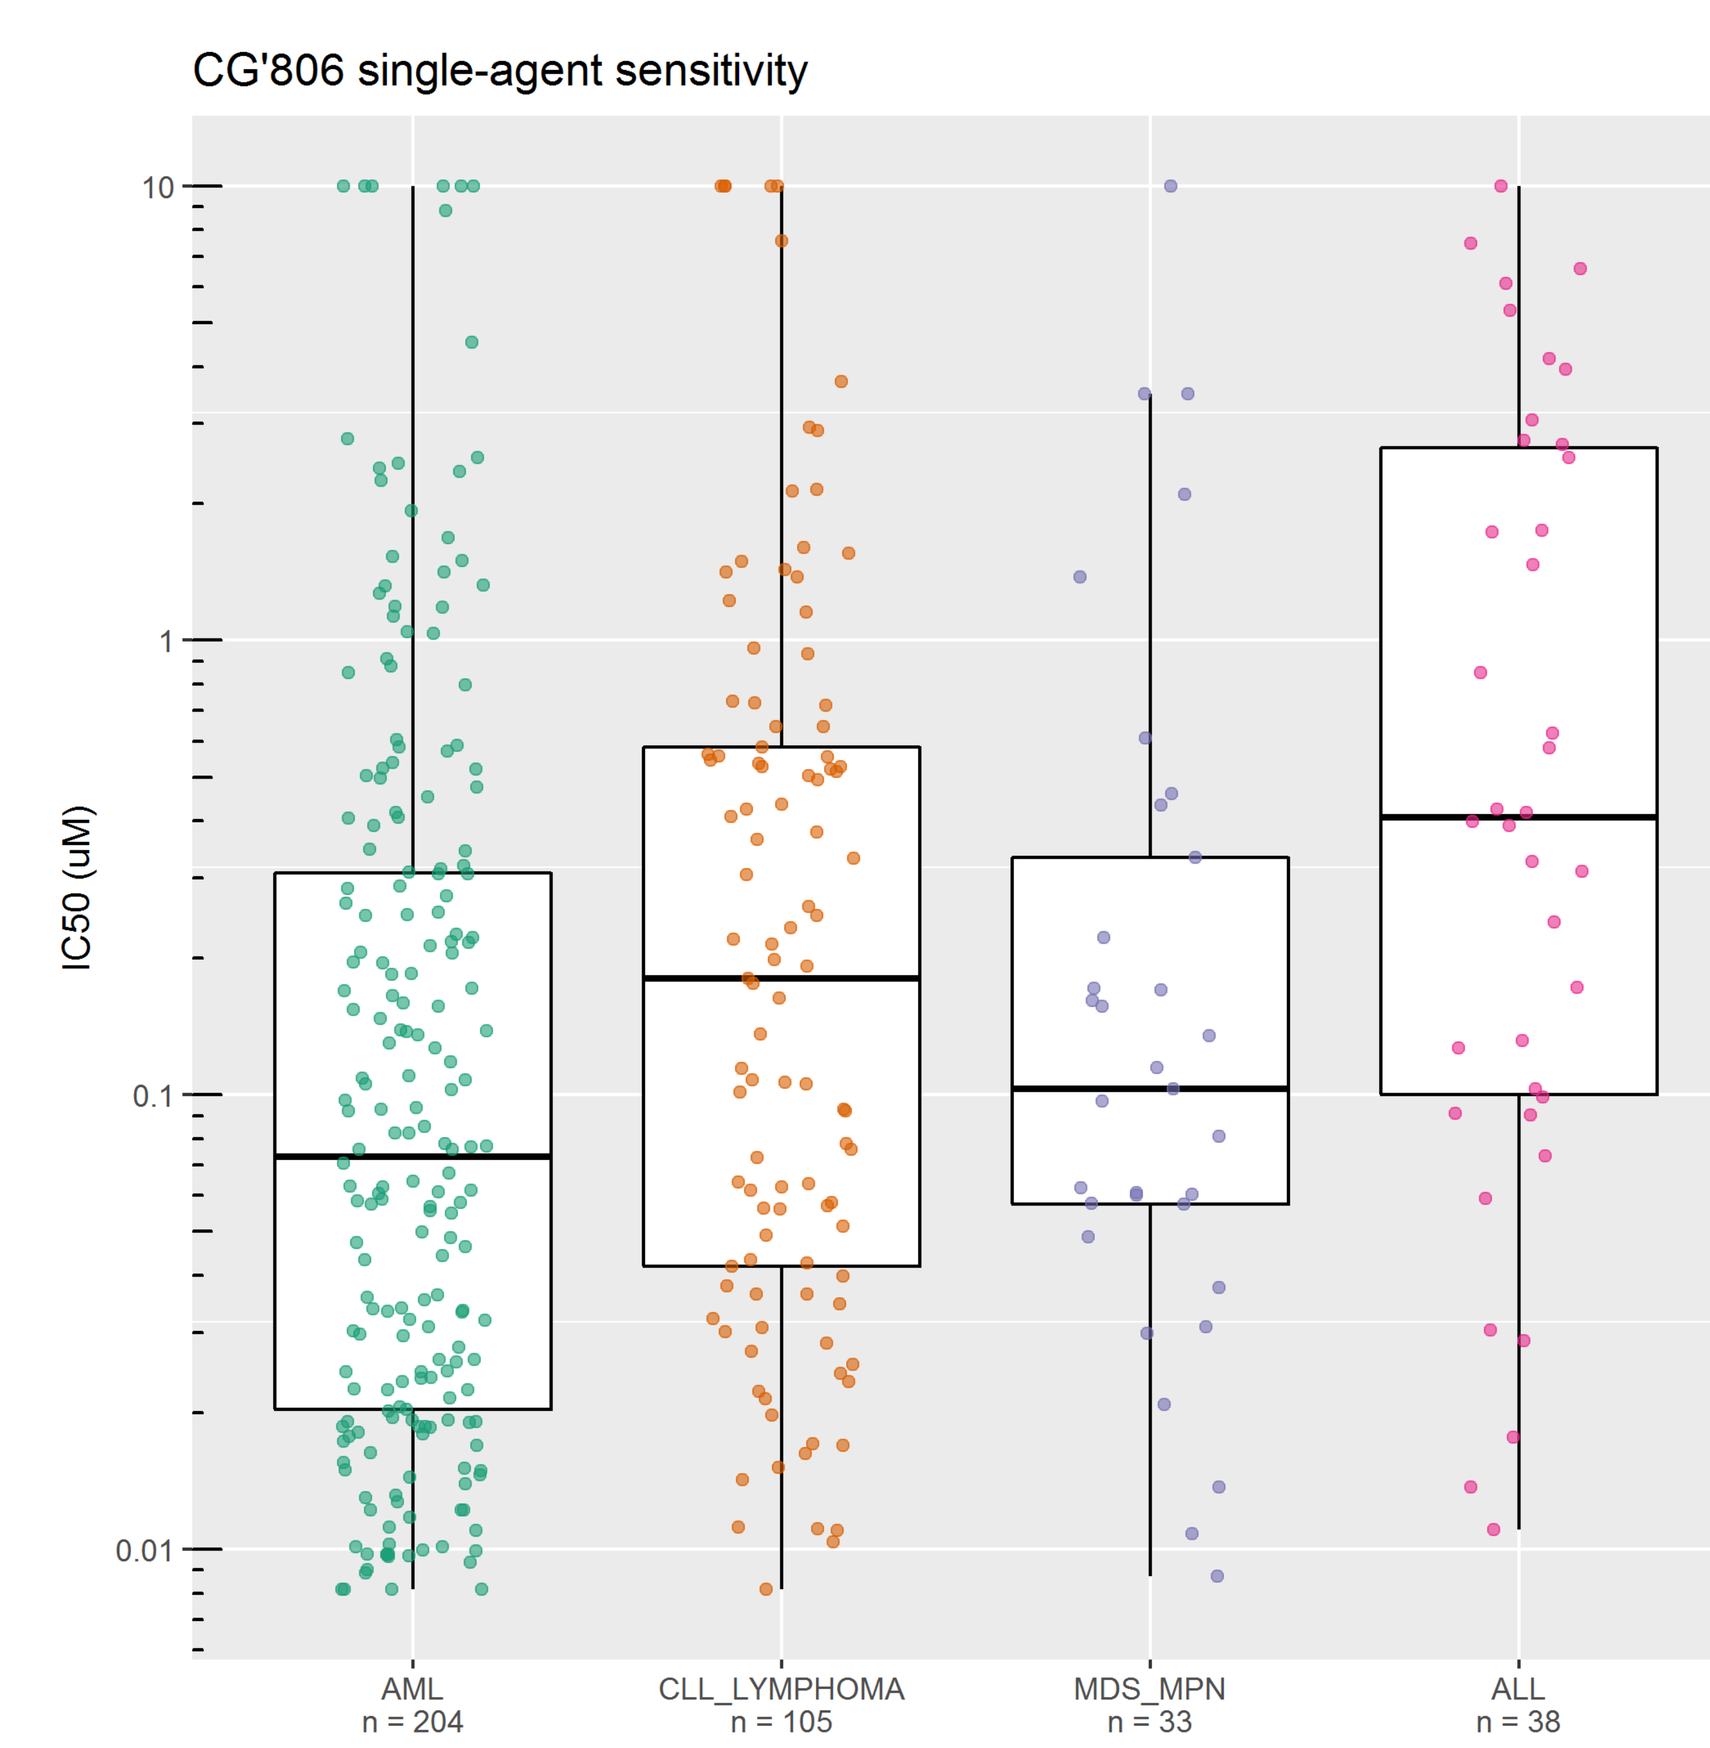
CG'806 single-agent sensitivity
IC50 (uM)
AML n = 204
CLL_LYMPHOMA n = 105
MDS_MPN n = 33
ALL n = 38

RESULTS

Across the four general subtypes of hematologic malignancies in the dataset with patient samples, there was broad sensitivity to CG’806, with 55% (90/164) AML, 48% (46/96) CLL, 22% (6/27) ALL, and 53% (14/26) MDS/MPN cases exhibiting an IC_{50} <0.1 μM. CG’806 demonstrated median IC_{50} values of 0.07 μM and 0.14 μM against primary AML and CLL cells, respectively. CG’806 also exerted potent picomolar to low-nanomolar IC_{50} anti-proliferative activity against human AML, B-ALL, mantle-cell lymphoma, Burkitt’s lymphoma, and diffuse large B-cell lymphoma cell lines.

CG’806 + OTX-105 combination

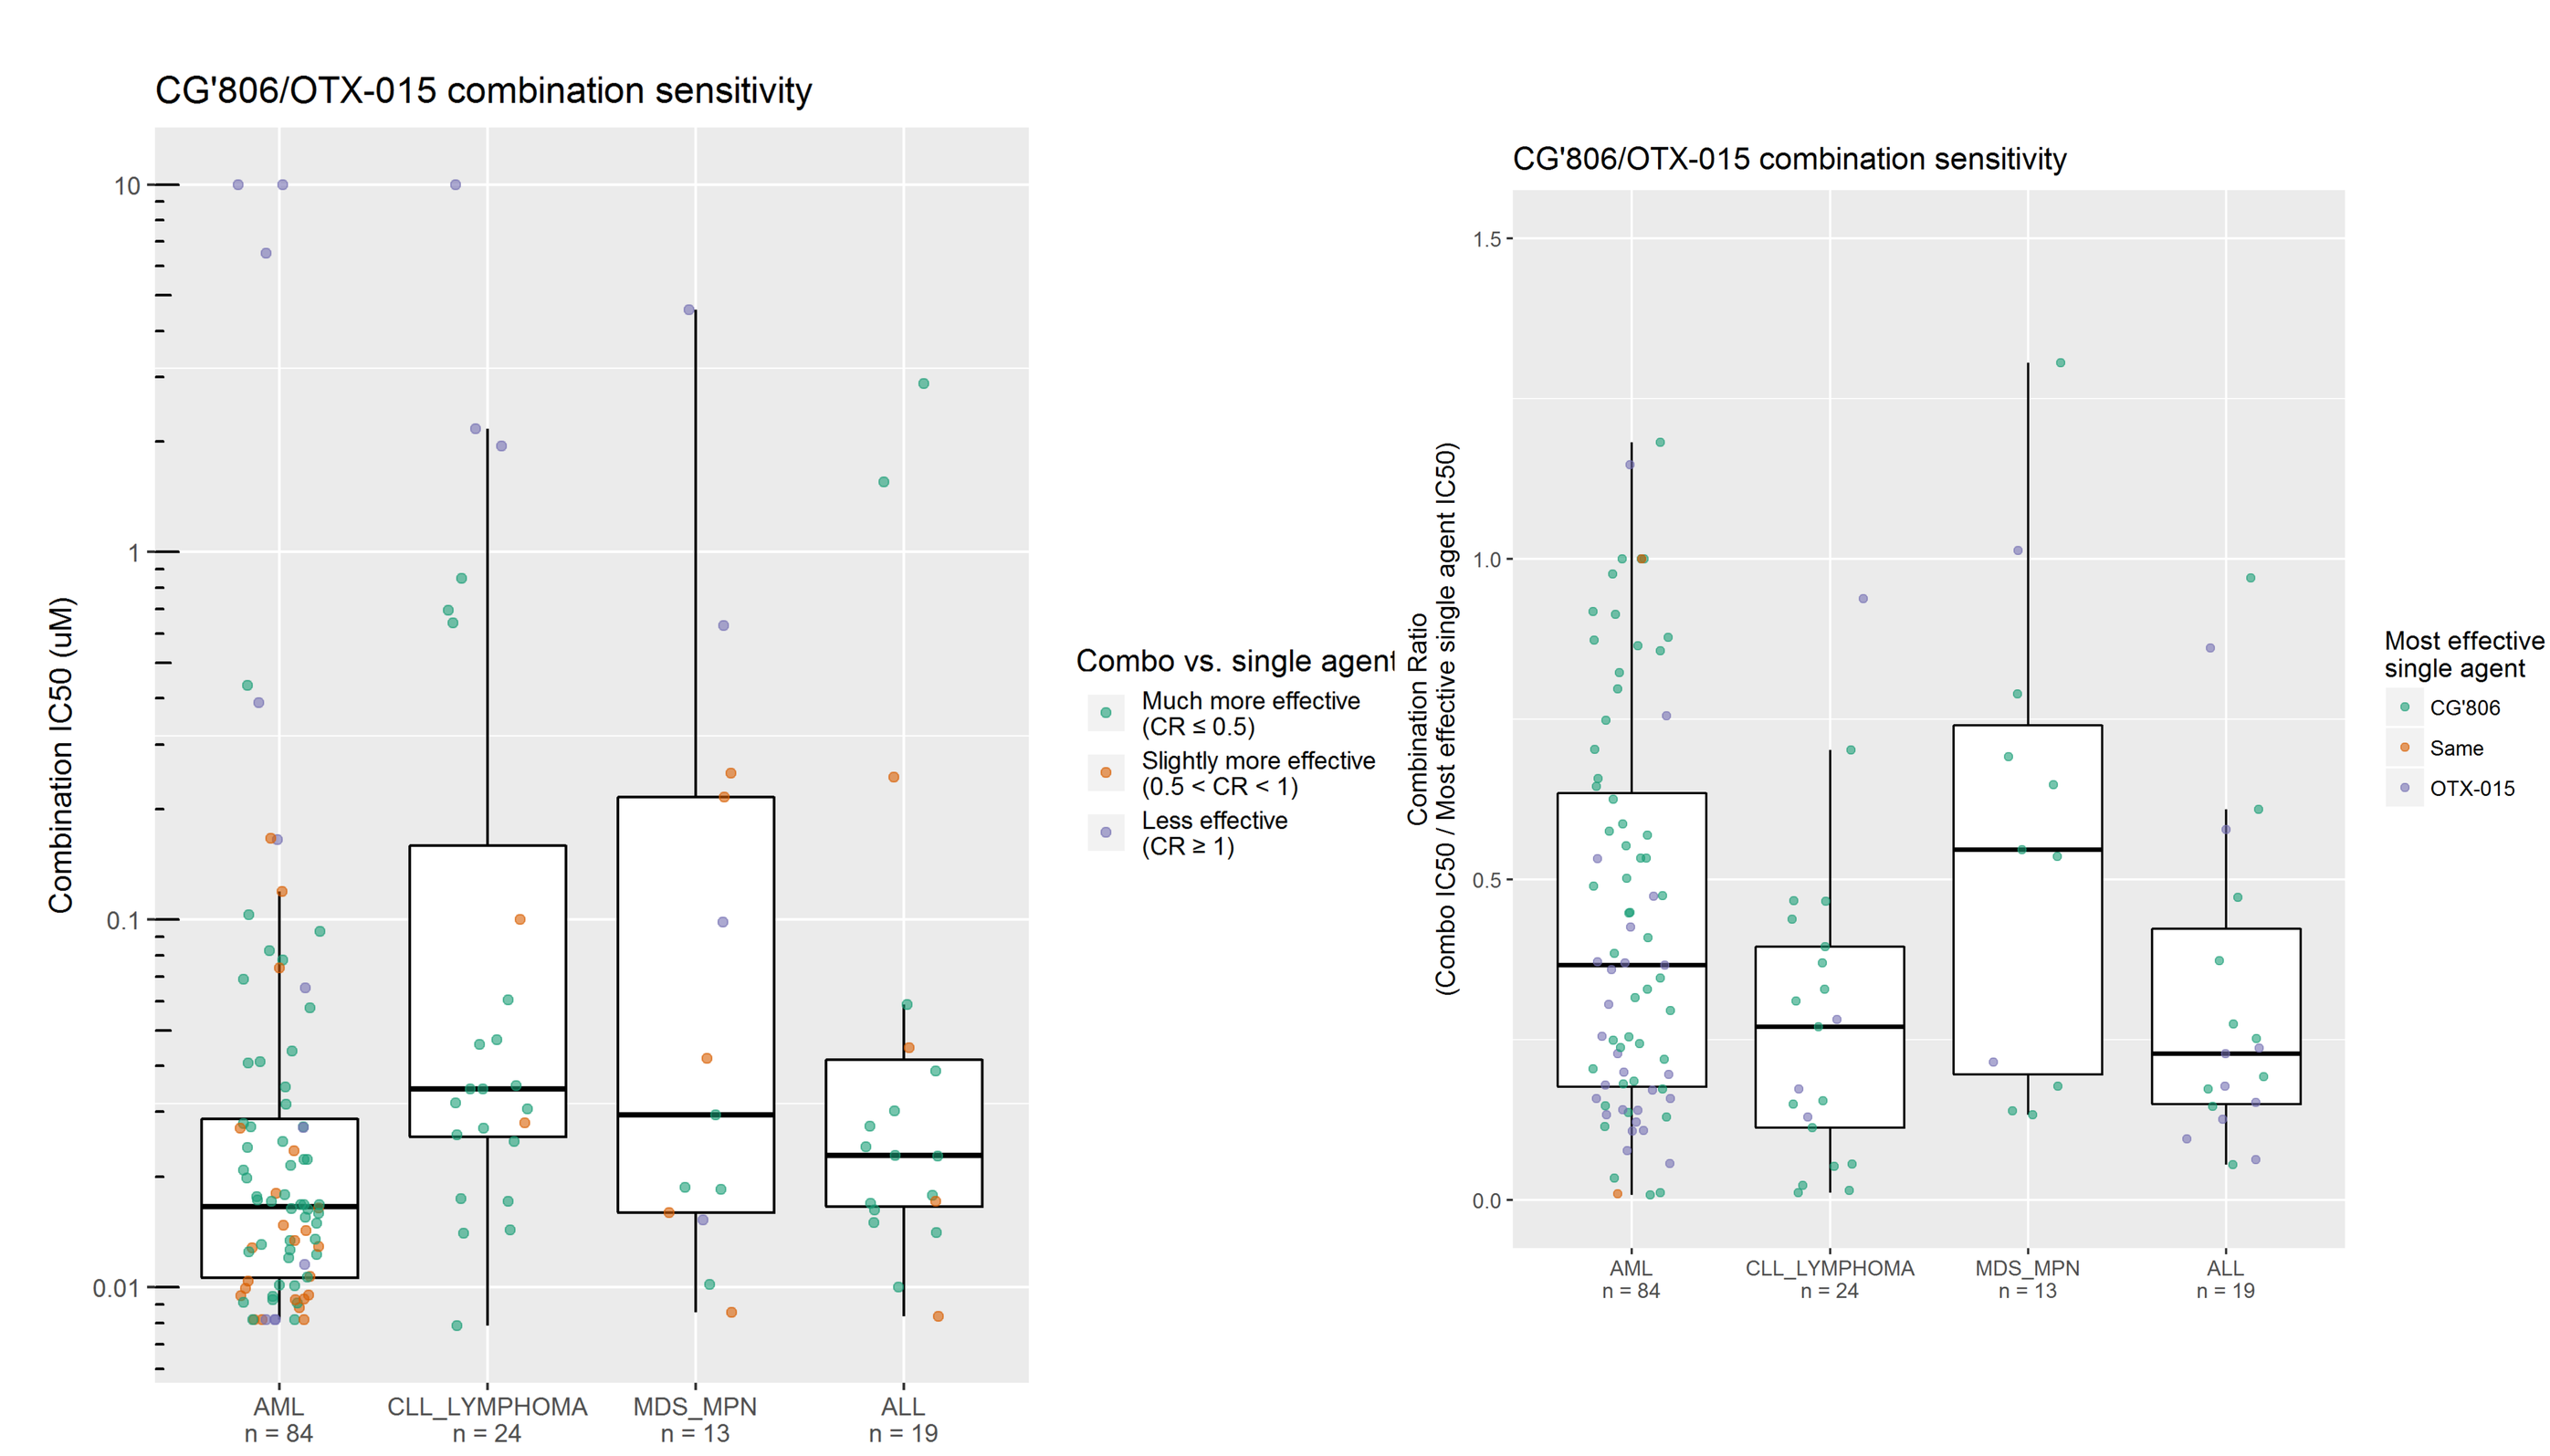
CG'806/OTX-015 combination sensitivity
Combination IC50 (uM)
Combo vs. single agent
Much more effective (CR ≤ 0.5)
Slightly more effective (0.5 < CR < 1)
Less effective (CR ≥ 1)
CG'806/OTX-015 combination sensitivity
Combination Ratio (Combo IC50 / Most effective single agent IC50)
Most effective single agent
CG'806
Same
OTX-015
AML n = 84
CLL_LYMPHOMA n = 24
MDS_MPN n = 13
ALL n = 19

CG’806 in combination with OTX-015 demonstrated median IC_{50} values of 0.02 μM and 0.04 μM against primary AML and CLL cells, respectively. In a majority of cases, the combination exhibited lower IC_{50} than both single-agents. These data illustrate that CG’806 enhances the activity of the OTX-015 BET bromodomain inhibitor against hematologic malignancy cells, and an analogous response was observed with CG’806 when combined with the JQ-1 BET bromodomain inhibitor (data not shown).

CG’806 + Venetoclax combination

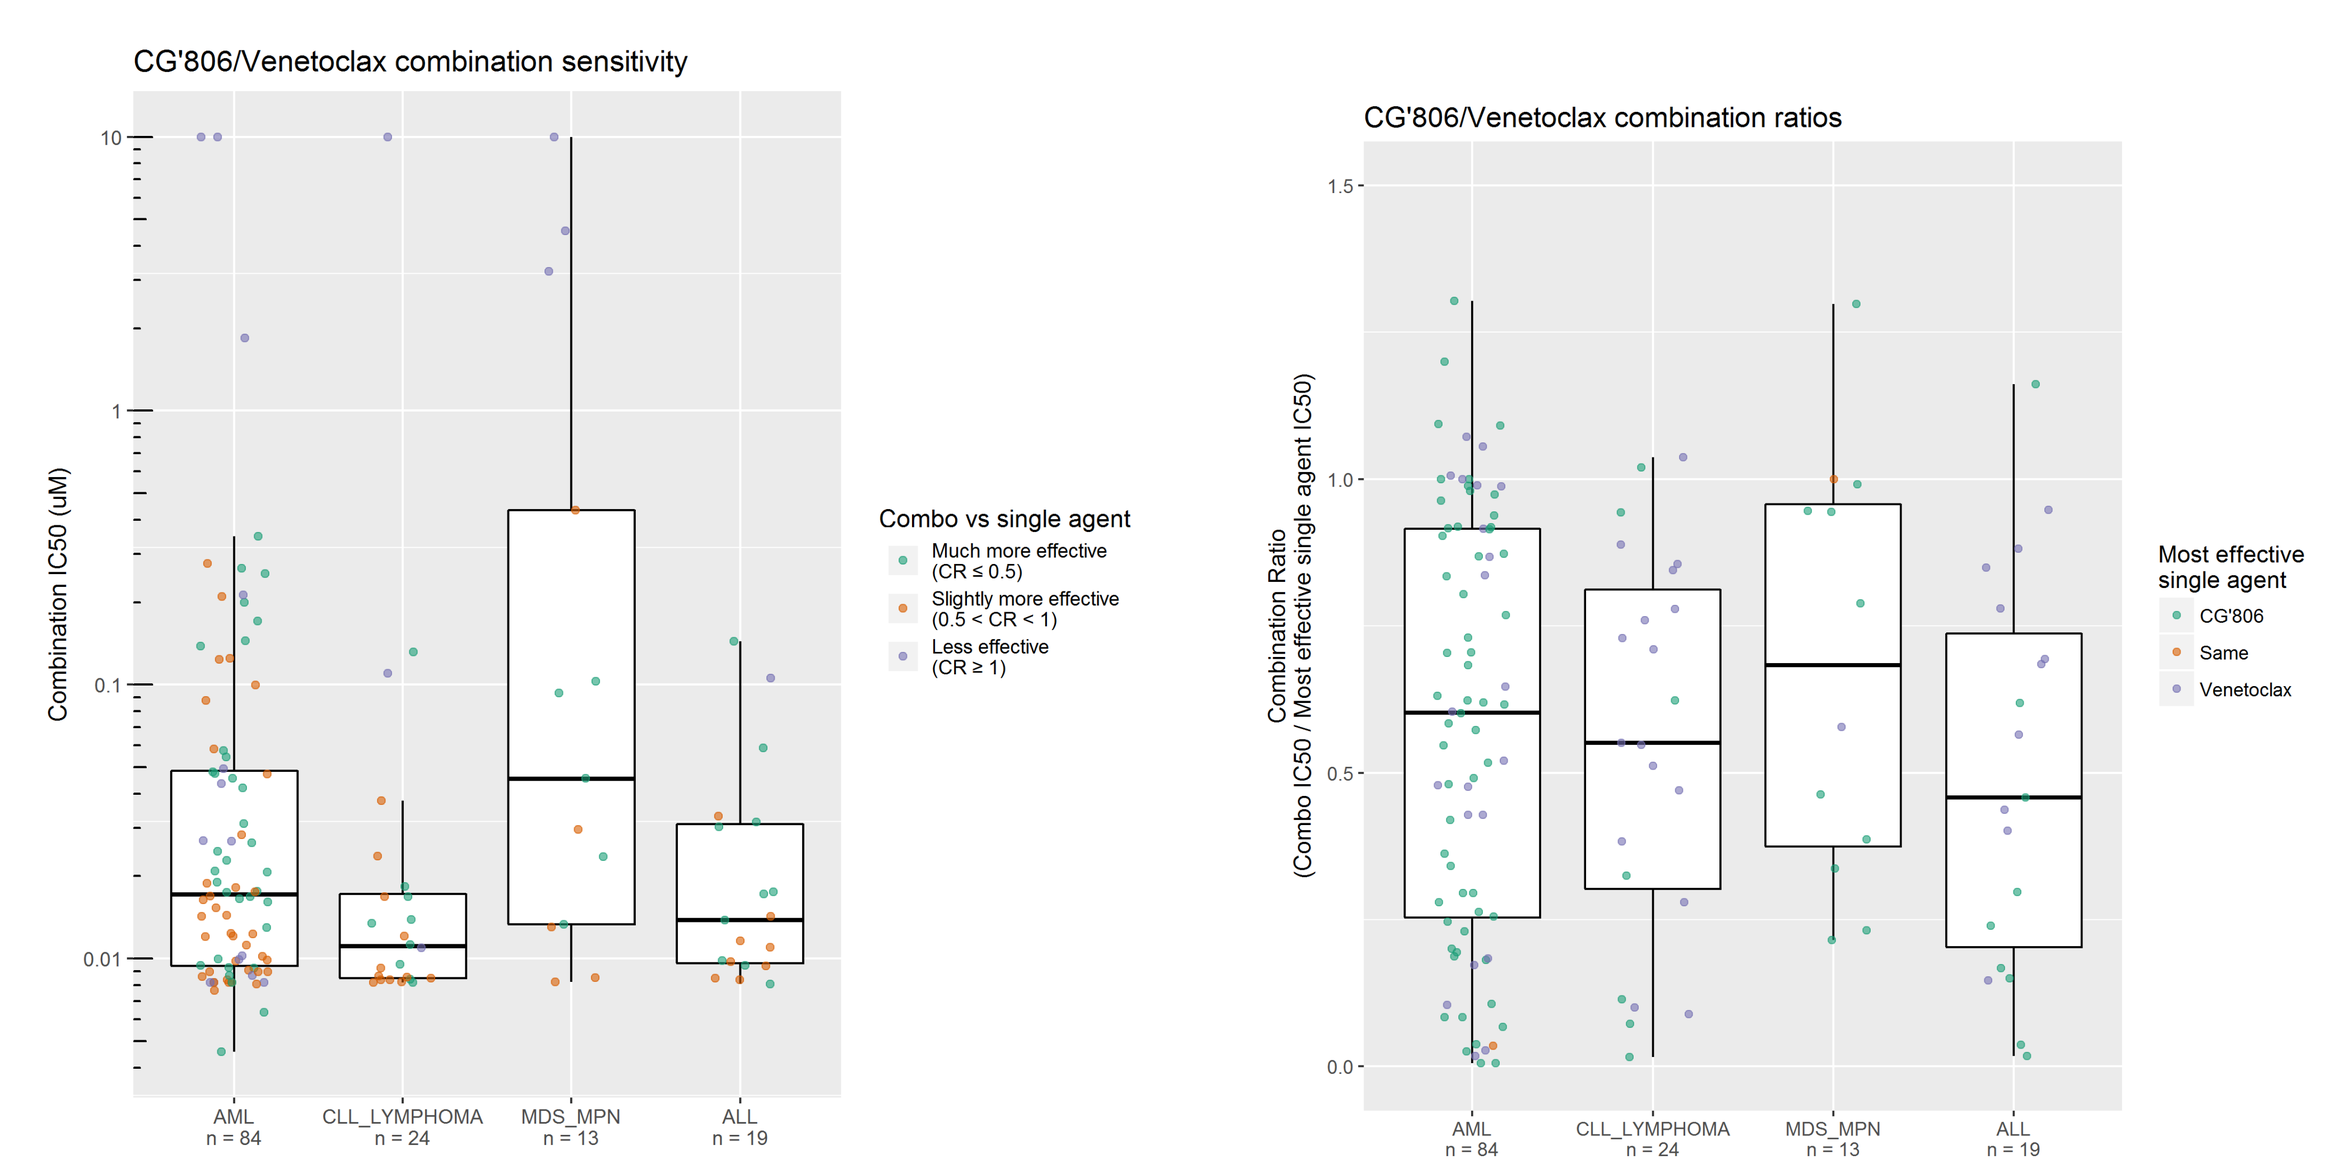
CG'806/Venetoclax combination sensitivity
Combination IC50 (uM)
Combo vs single agent
Much more effective (CR ≤ 0.5)
Slightly more effective (0.5 < CR < 1)
Less effective (CR ≥ 1)
CG'806/Venetoclax combination ratios
Combination Ratio (Combo IC50 / Most effective single agent IC50)
Most effective single agent
CG'806
Same
Venetoclax
AML n = 84
CLL_LYMPHOMA n = 24
MDS_MPN n = 13
ALL n = 19

CG’806 in combination with venetoclax demonstrated median IC_{50} values of 0.02 μM and 0.01 μM against primary AML and CLL cells, respectively. In a majority of cases, the combination exhibited lower IC_{50} than both single-agents. These data demonstrate the benefit of simultaneously targeting specific clusters of kinases by CG’806 and the Bcl-2 anti-apoptotic protein by venetoclax.

CG’806 Induces Apoptosis

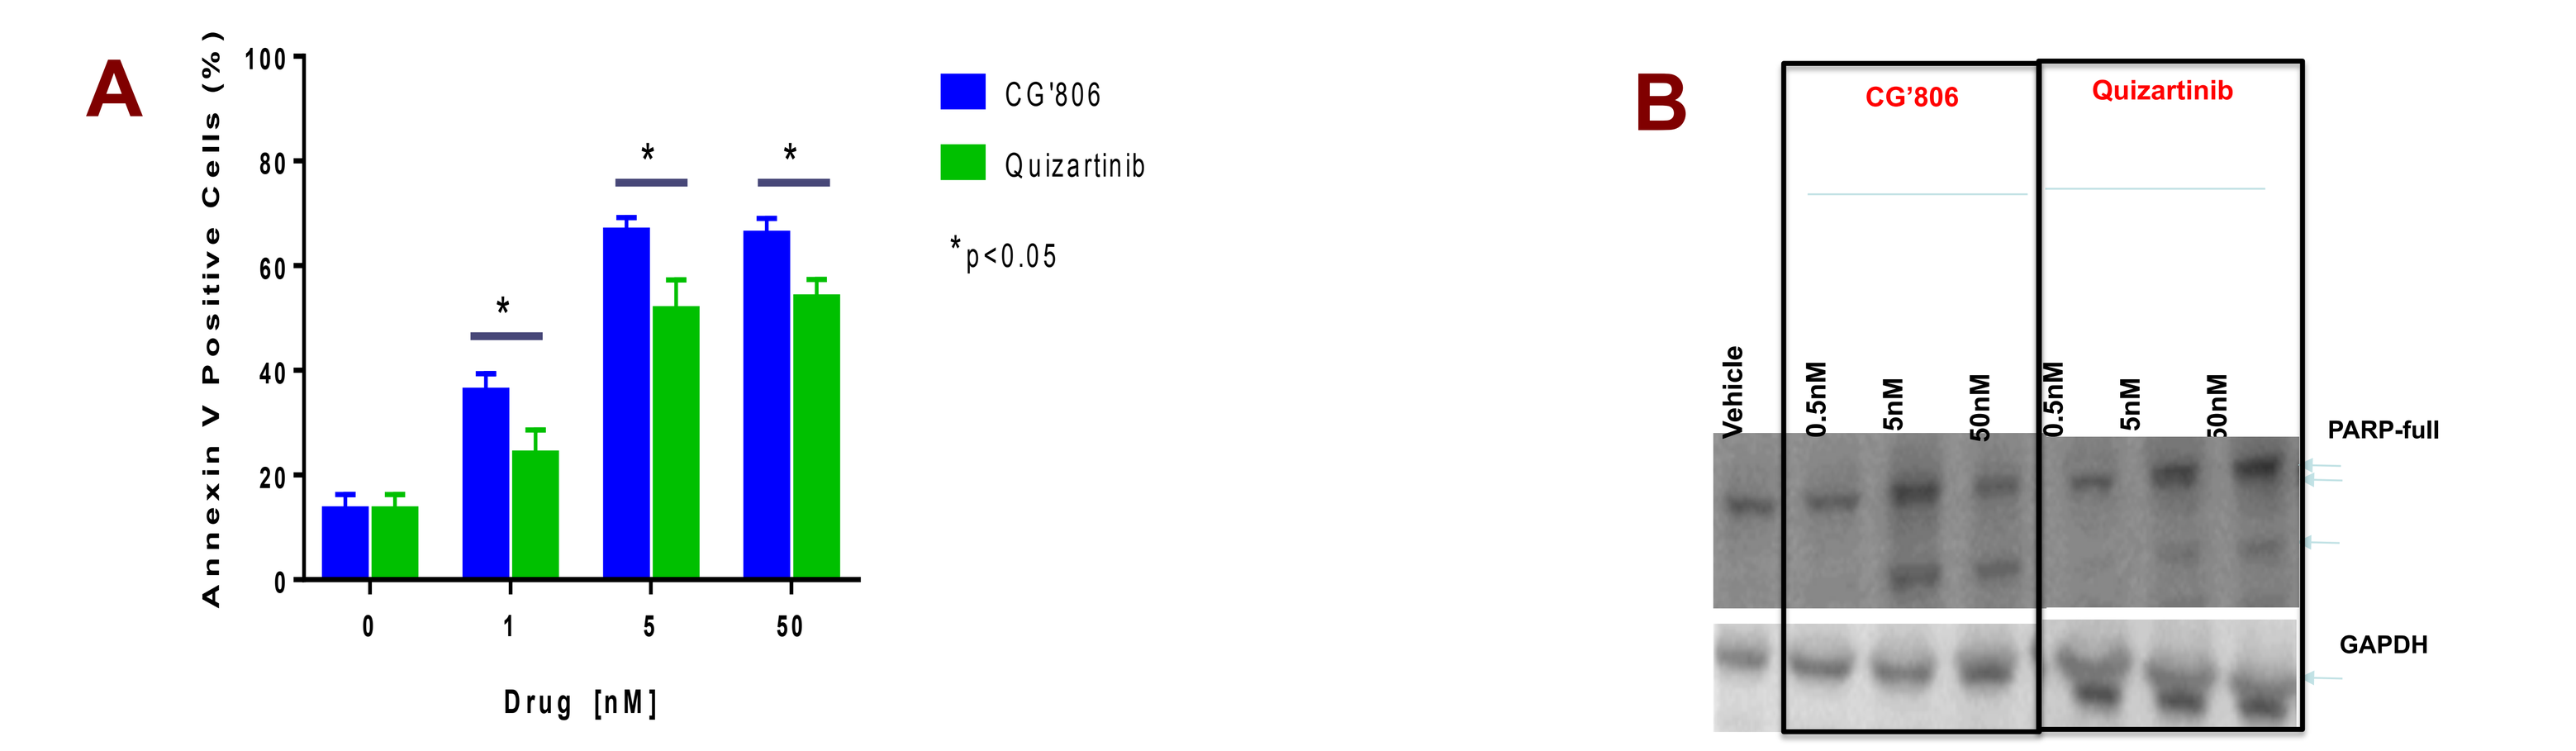
A
Annexin V Positive Cells (%)
Drug [nM]
CG'806
Quizartinib
*p<0.05
B
CG'806
Quizartinib
Vehicle
0.5nM
5nM
50nM
PARP-full
GAPDH

CG’806 induces apoptosis in a FLT3-ITD positive cell line MV4-11 more effectively than quizartinib.

A. MV4-11 cells were treated with vehicle DMSO, CG’806 or quizartinib at indicated concentration for 24 hour and then subjected to Annexin V-FITC and PI staining for flow cytometry analysis (n=3).

B. Western Blotting on full- and cleaved- PARP signals (representative blots of three independent experiments).

CG’806 Inhibits Signaling through FLT3

CG’806 inhibited FLT3 signaling more effectively than does a FLT3 inhibitor quizartinib. MV4-11 cells were treated with vehicle DMSO, CG’806 or quizartinib at indicated concentration for 1 hour and then subjected to Western Blotting on phosphor-FLT3 and its phosphor-ERK and phosphor-STAT downstream signaling pathways (representative blots of three independent experiments).

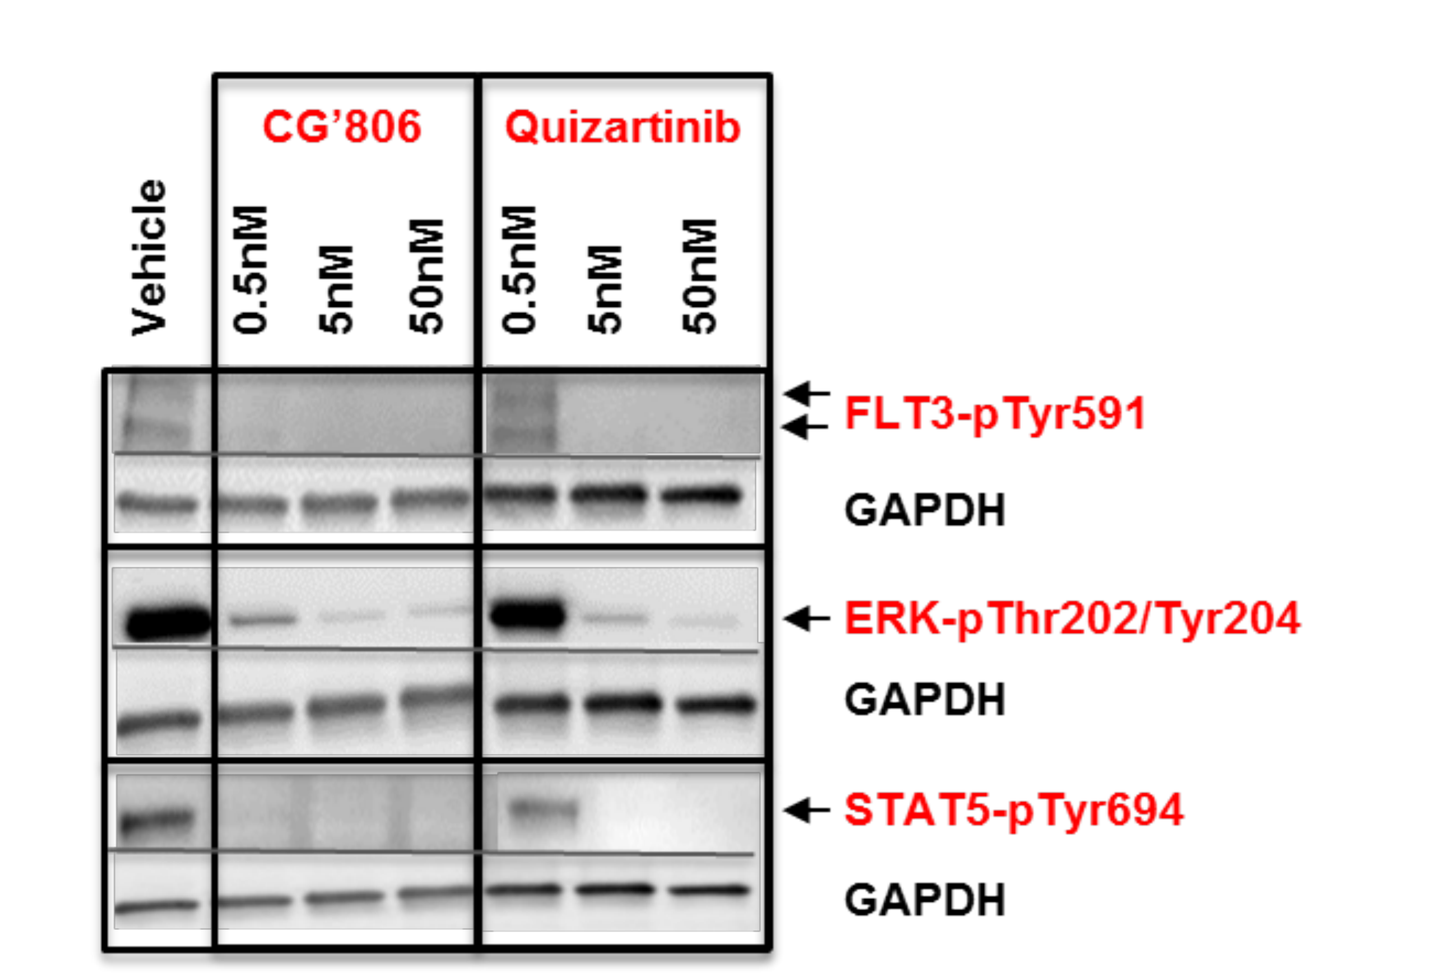
CG'806
Quizartinib
Vehicle
0.5nM
5nM
50nM
FLT3-pTyr591
GAPDH
ERK-pThr202/Tyr204
GAPDH
STAT5-pTyr694
GAPDH

CONCLUSIONS

- CG’806 exerts potent cytotoxic activity on primary samples from patients with AML, CLL and other hematologic malignancies, as well as and cultured cell lines.
- CG’806 induces apoptosis in FLT3-ITD AML cell lines more potently than the quizartinib FLT3-ITD inhibitor.
- Combinations of CG’806 with a BET bromodomain inhibitor or a BCL2 inhibitor show enhanced effectiveness, suggesting these classes of drugs are potential combination partners for CG’806.
- The broad range of activity of CG’806 against hematologic malignancy cells results from its ability to inhibit multiple oncogenic pathways, supporting further development of this CG’806 for AML, CLL and other hematologic malignancies.