VIAGENPUMATUCEL-L (HS-110) IN COMBINATION WITH NIVOLUMAB IN PREVIOUSLY-TREATED PATIENTS WITH ADVANCED NON-SMALL LUNG CANCER (NSCLC)

Daniel Morgensztern[1], Lyudmila Bazhenova[2], Saiama N. Waqar[1], Lori McDermott[3], Jeff Hutchins[3], Wael Harb[4], Nathan Pennell[5], Roger B. Cohen[6]

[1]Washington University School of Medicine, St. Louis, MO; [2]UC San Diego, Moores Cancer Center, San Diego, CA; [3]Heat Biologics, Inc, Durham, NC; [4]Horizon Oncology Center, Lafayette, IN; [5]Taussig Cancer Institute, Cleveland Clinic, Cleveland, OH; [6]University of Pennsylvania Perelman School of Medicine, Philadelphia, PA

Background

Viagenpumatucel-L (HS-110) is an allogeneic cellular immunotherapy that incorporates a broad range of tumor antigens that are known to be shared amongst a high proportion of patients with non-small cell lung cancer (NSCLC). This cell system contains gp96-Ig and is designed to enable the cell to express gp96 in secreted form. The secreted gp96 acts as a chaperone to induce cellular immune responses to the tumor antigens expressed by **Viagenpumatucel-L (HS-110)**. gp96 is a unique chaperone because it can activate MHC and up-regulate T-cell co-stimulation and deliver chaperoned antigens to an APC for display via MHC I, with the net result being CD8+ T-cell mediated immune responses[1,2].

The HS110-102 "Durga" Trial is an exploratory, multi-cohort master protocol evaluating HS-110 in combination with anti-PD1 mAbs in the treatment of advanced non-small lung cancer. Here we present top line data from Cohort A. This cohort is comprised of previously-treated patients who have not received a checkpoint inhibitor (CPI) prior to study entry. NCT Trial ID: **NCT02439450**

Mechanism of Action

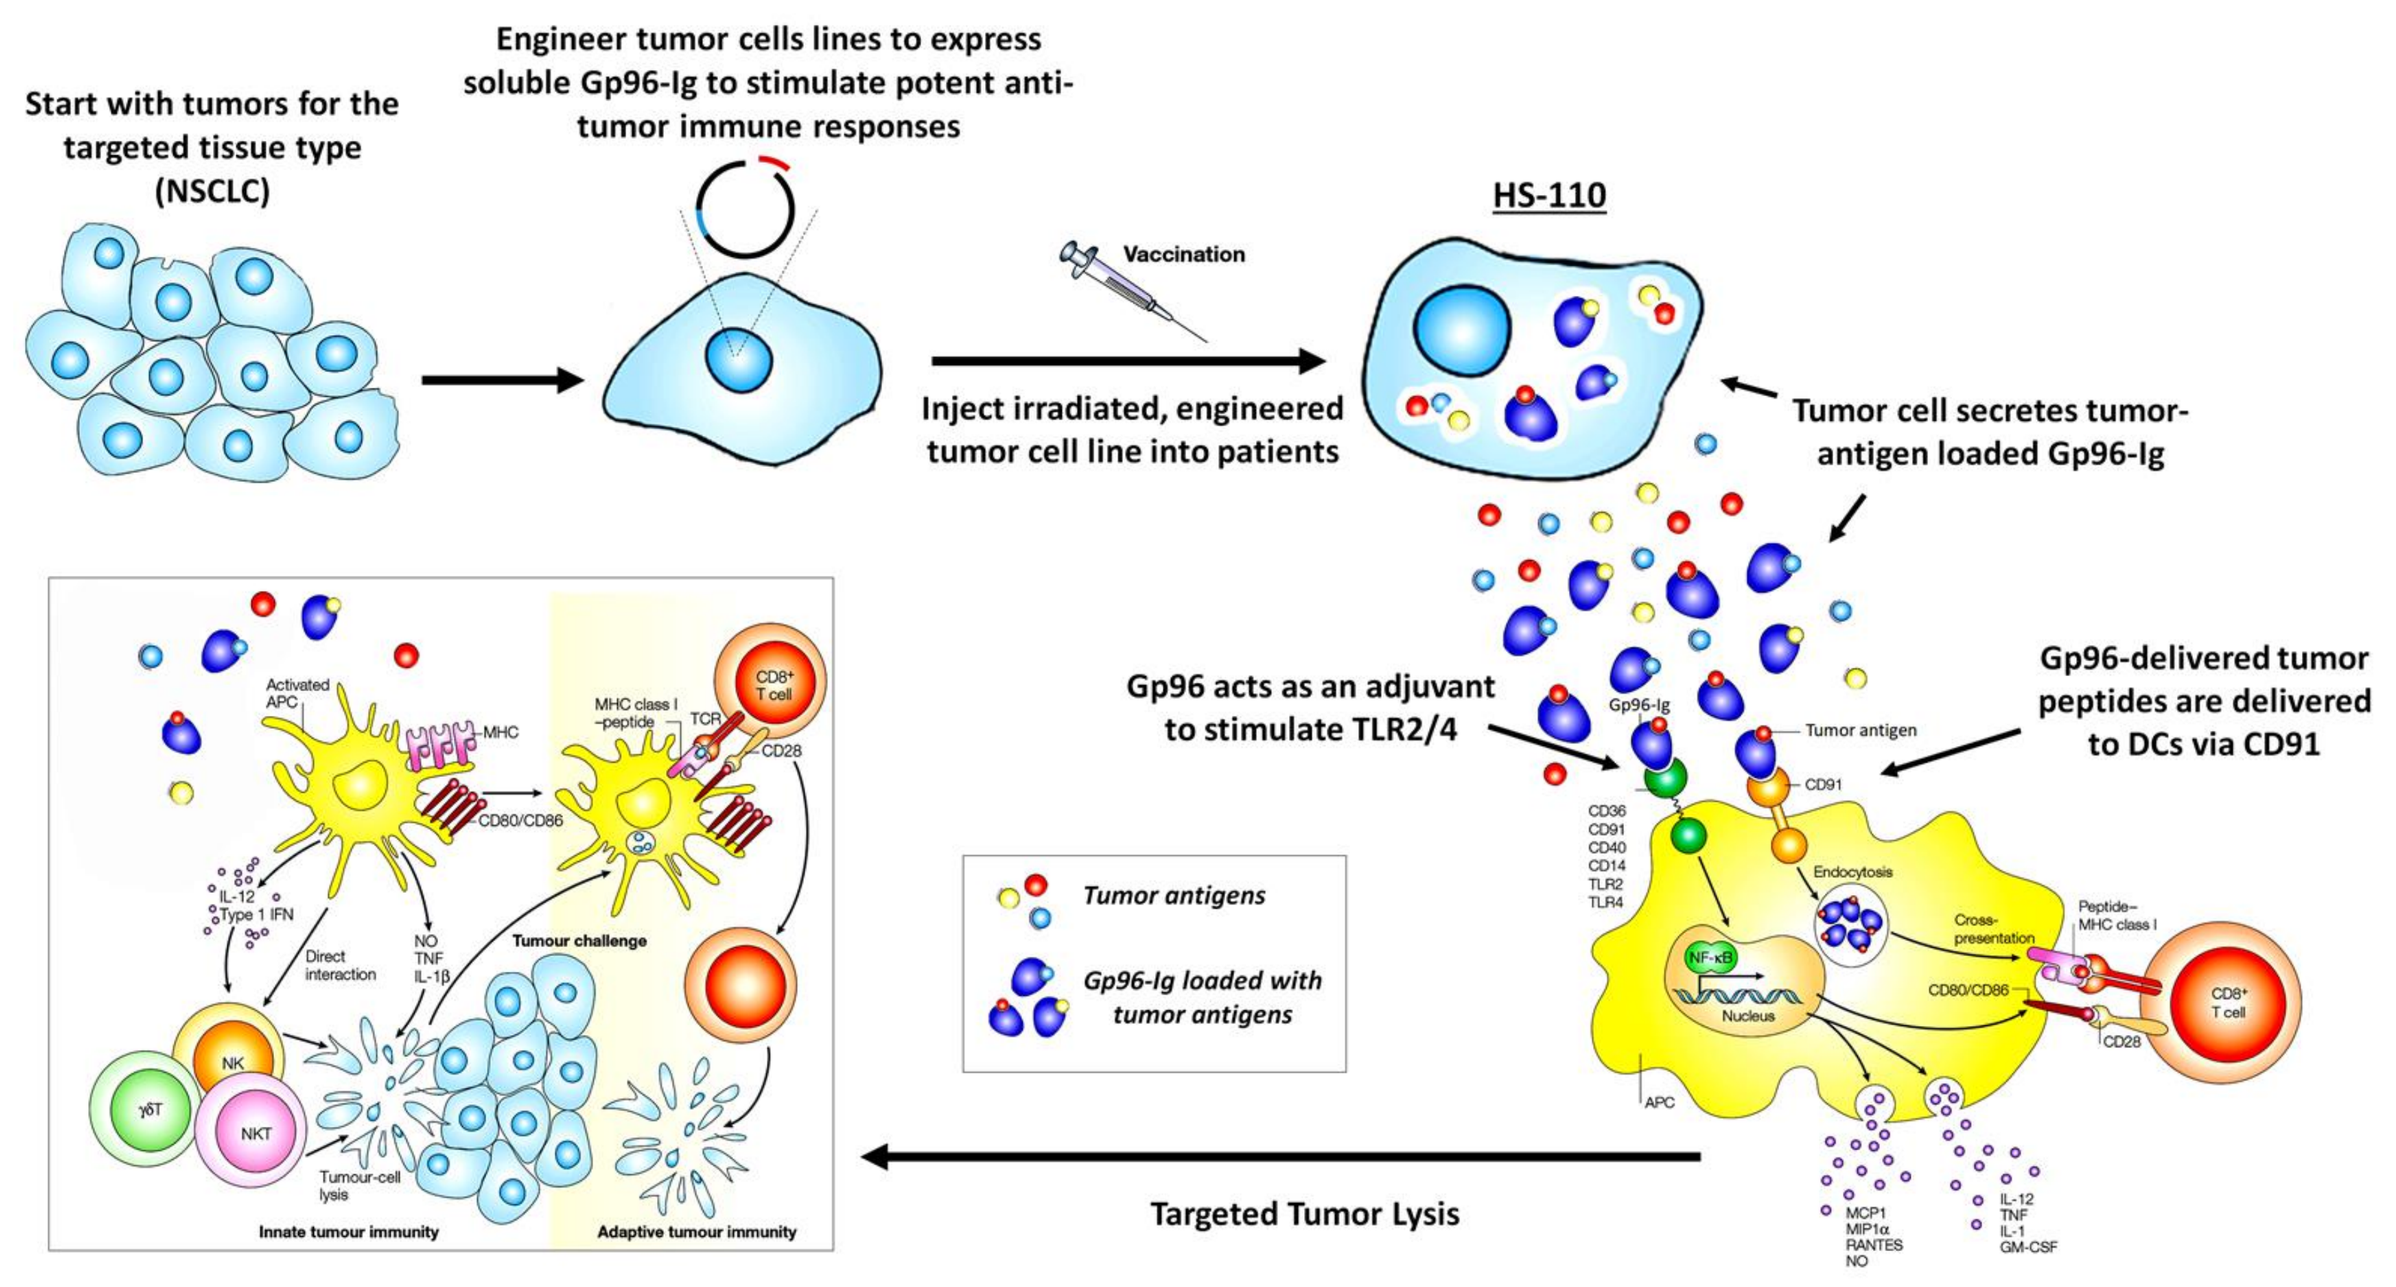
Start with tumors for the targeted tissue type (NSCLC)
Engineer tumor cells lines to express soluble Gp96-Ig to stimulate potent anti-tumor immune responses
Vaccination
Inject irradiated, engineered tumor cell line into patients
HS-110
Tumor cell secretes tumor-antigen loaded Gp96-Ig
Gp96 acts as an adjuvant to stimulate TLR2/4
Gp96-delivered tumor peptides are delivered to DCs via CD91
Tumor antigens
Gp96-Ig loaded with tumor antigens
Targeted Tumor Lysis

Figure 1: Viagenpumatucel-L (HS-110) Mechanism of Action and Pre-clinical Activity
HS-110 is derived from a lung adenocarcinoma cell line transfected with gp96-Ig, which acts as a chaperone protein for tumor associated antigens and is recognized by CD91 on APCs, resulting in cross-presentation of antigen to MHC I for the selection of antigen-specific CD8 cells. Gp96-Ig also binds to TLRs 2 and 4 leading to upregulation of co-stimulatory molecules, which include MHC II and secretion of cytokines and chemokines.

Study Schema

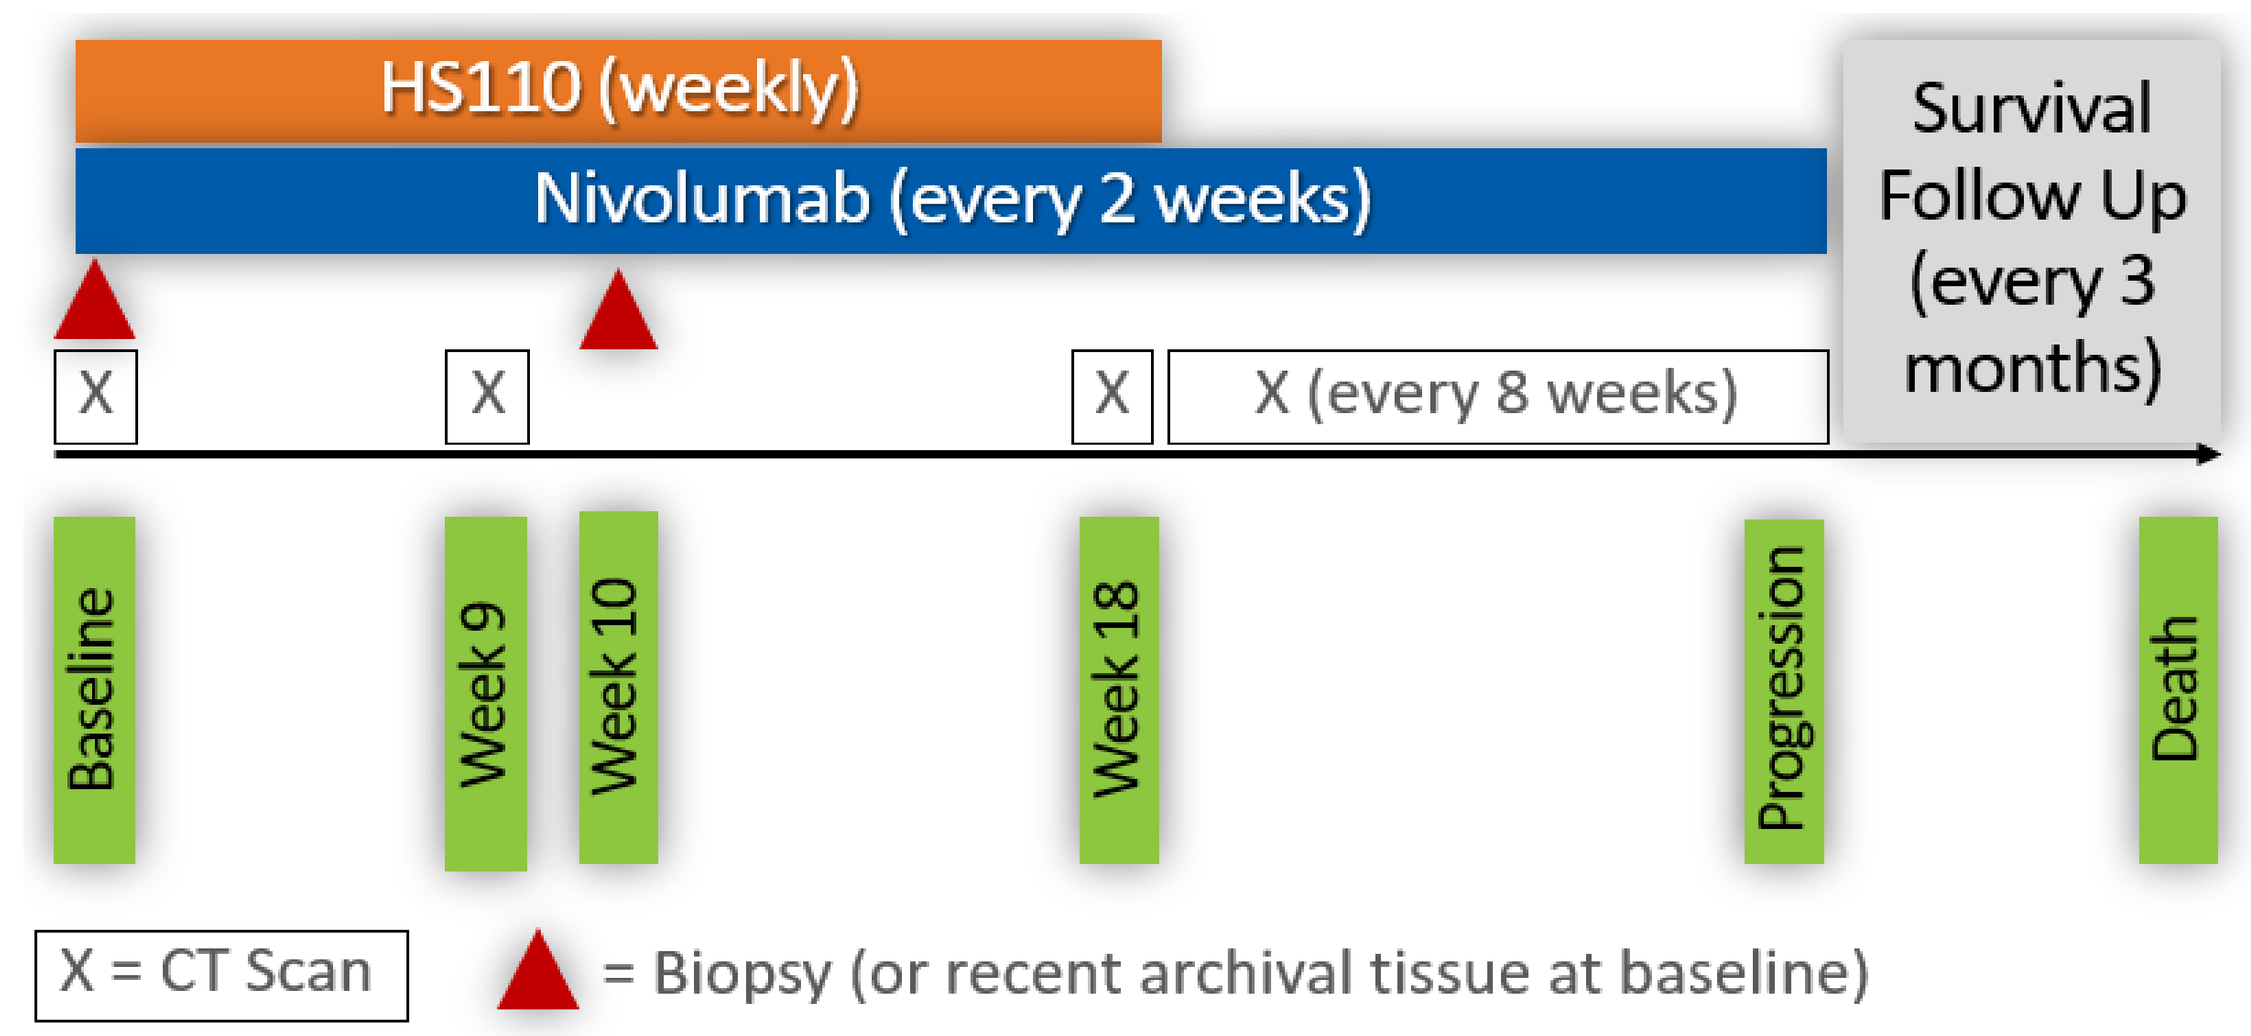
HS110 (weekly)
Nivolumab (every 2 weeks)
Survival Follow Up (every 3 months)
X
X
X
X (every 8 weeks)
Baseline
Week 9
Week 10
Week 18
Progression
Death
X = CT Scan
= Biopsy (or recent archival tissue at baseline)

Figure 2: HS110-102 Study Schema
Patients receive weekly HS-110 (1 x 10^7 cells) intradermally for 18 weeks via 5 simultaneous injections of 0.1ml each, and biweekly nivolumab 240 mg IV until disease progression or unacceptable toxicity.

Patient Characteristics

		ITT (N = 46)
Median age (range)		65 (37 – 87)
Female gender		26 (57%)
Caucasian		41 (89%)
ECOG PS 1		32 (70%)
EGFR or ALK positive		6 (13%)
Histology	Adeno Squamous	43 (93%) 3 (7%)
Smoking status	Current/past Never	39 (85%) 7 (15%)
Prior lines of tx	1 2 3 or more	31 (67%) 7 (15%) 8 (18%)
PD-L1	< 1% ≥ 1% Unevaluable	21 (46%) 9 (19%) 16 (35%)

Table 1: Patient Characteristics
Baseline patient demographics of Intent-to-treat population (n=46).

Best Overall Response

	RECIST 1.1	iRECIST
ORR	**20% (9)**	**22% (10)**
PR	20% (9)	22% (10)
SD	26% (12)	26% (12)
Not Evaluable	7% (15)	7% (15)
DCR	**46% (21)**	**48% (22)**

Table 2: Objective Response Rates
ORR of the Intent-to-treat population (n=46) performed locally by study Investigators using RECIST 1.1. iRECIST shown as one patient achieved confirmed PR after initial radiographic PD.

T Cell Changes by Best Overall Response (BOR)

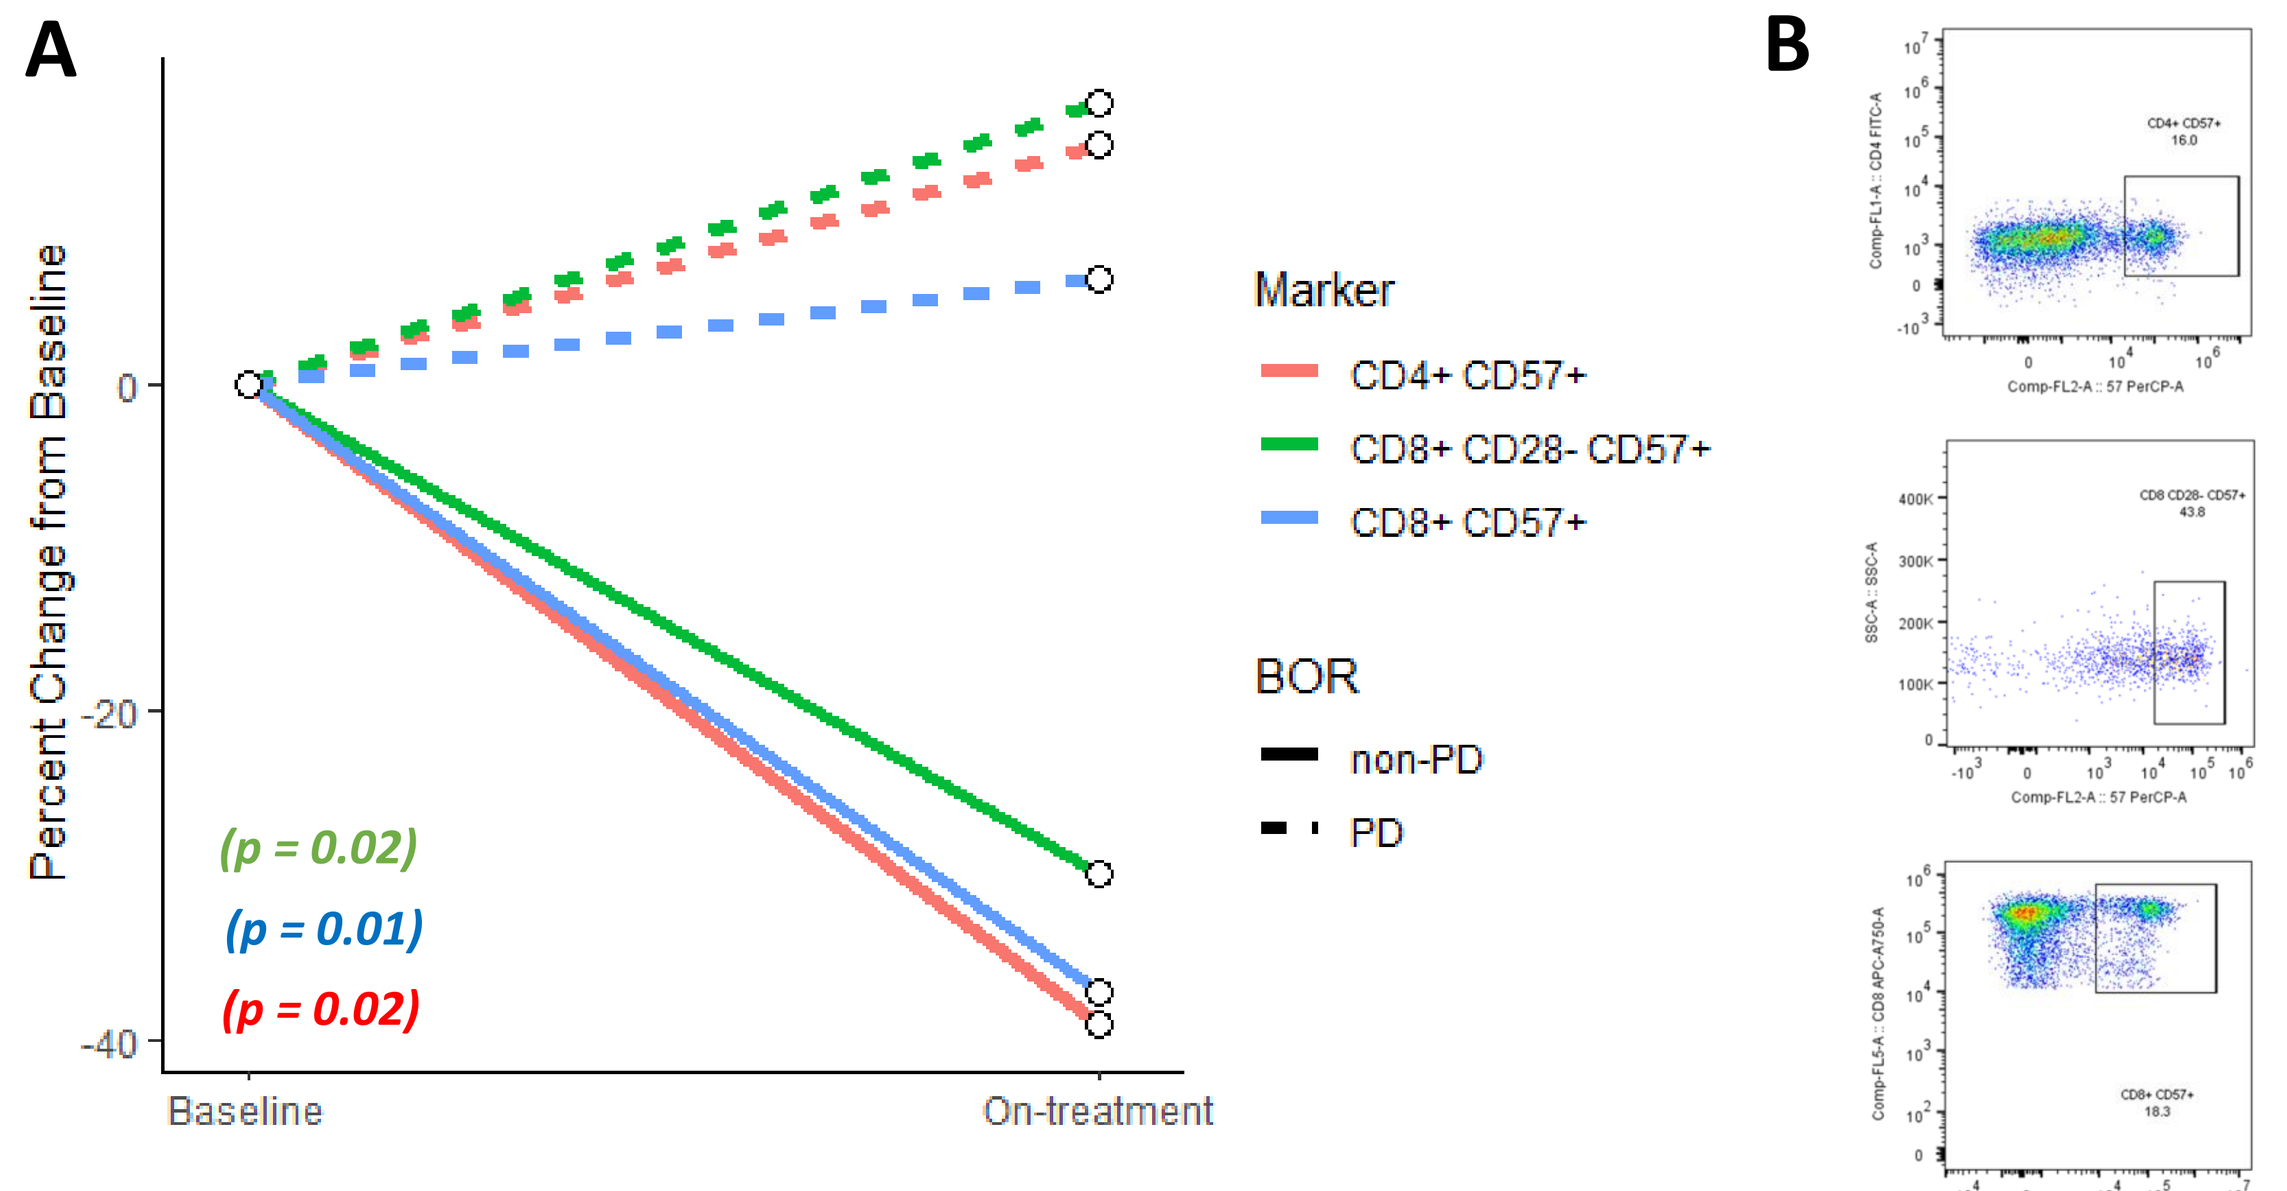
A
B
Percent Change from Baseline
0
-20
-40
Baseline
On-treatment
(p = 0.02)
(p = 0.01)
(p = 0.02)
Marker
CD4+ CD57+
CD8+ CD28- CD57+
CD8+ CD57+
BOR
non-PD
PD

Figure 3: T Cell Subset Changes in Peripheral Blood Based on BOR
A. Mean flow cytometric measurements of T cell subset changes in 22 patients at baseline and on-treatment based on CD57+ (terminal differentiation) expressed on CD8+ (effector), CD8+CD28- (effector memory) and CD4+ (helper) cells according to the patient's BOR clinical outcome by RECIST 1.1: Progressive Disease (PD, n=9) or non-Progressive Disease (SD or PR, n=13). This downward trend is an indicator of effective immunity in an antigen driven population of effector cells. **B.** Representative flow cytometry histograms of peripheral blood T cell subpopulations for a study patient at baseline.

Overall Survival

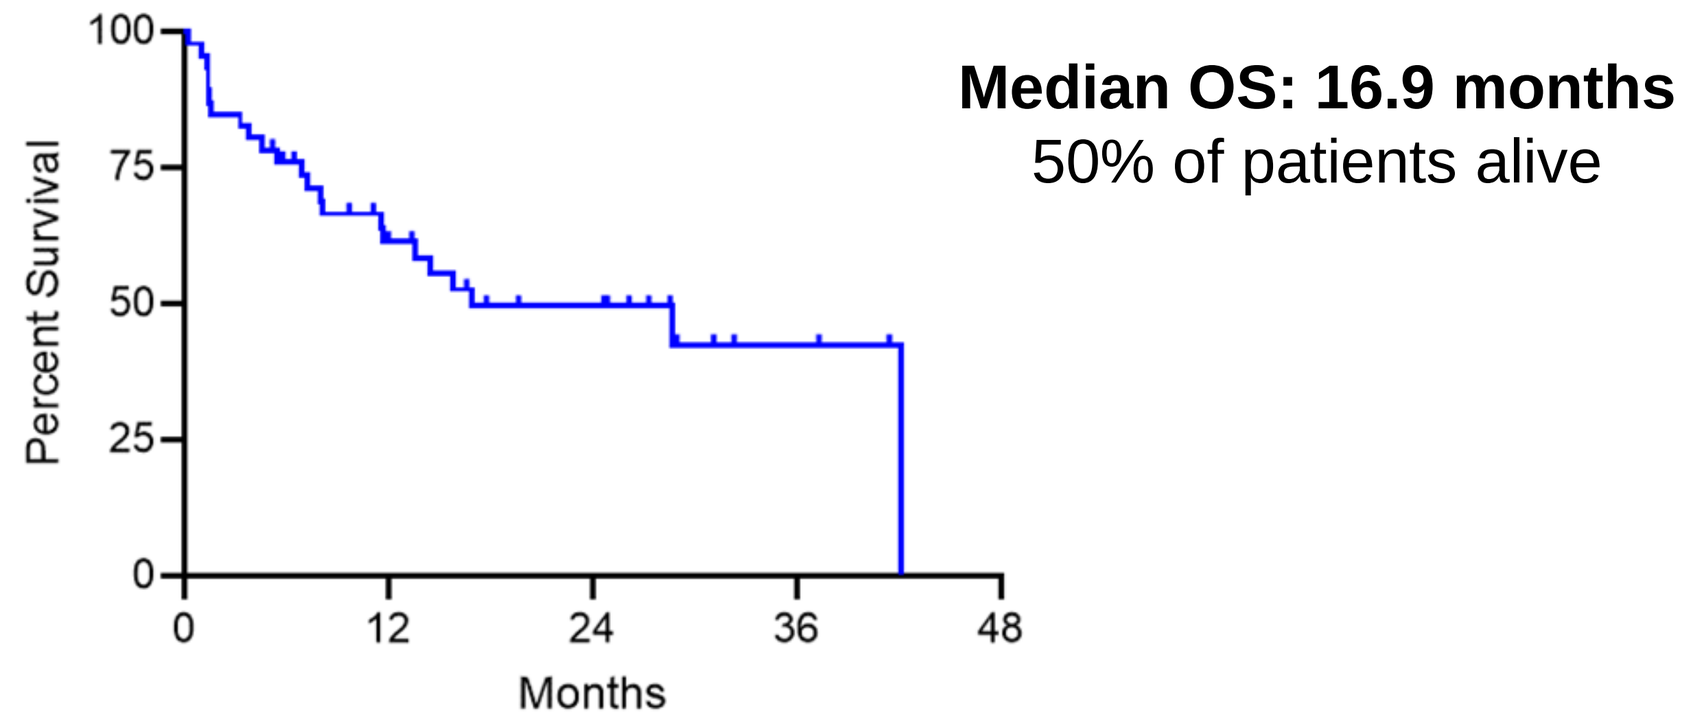
Median OS: 16.9 months
50% of patients alive
Percent Survival
Months

Figure 4: Kaplan Meier of estimated Overall Survival – ITT Population
Overall survival of ITT population (N=46). Twenty-three (23) patients censored. mOS is estimated by KM to be 16.9 months [95% CI; 11.6, 42.1].

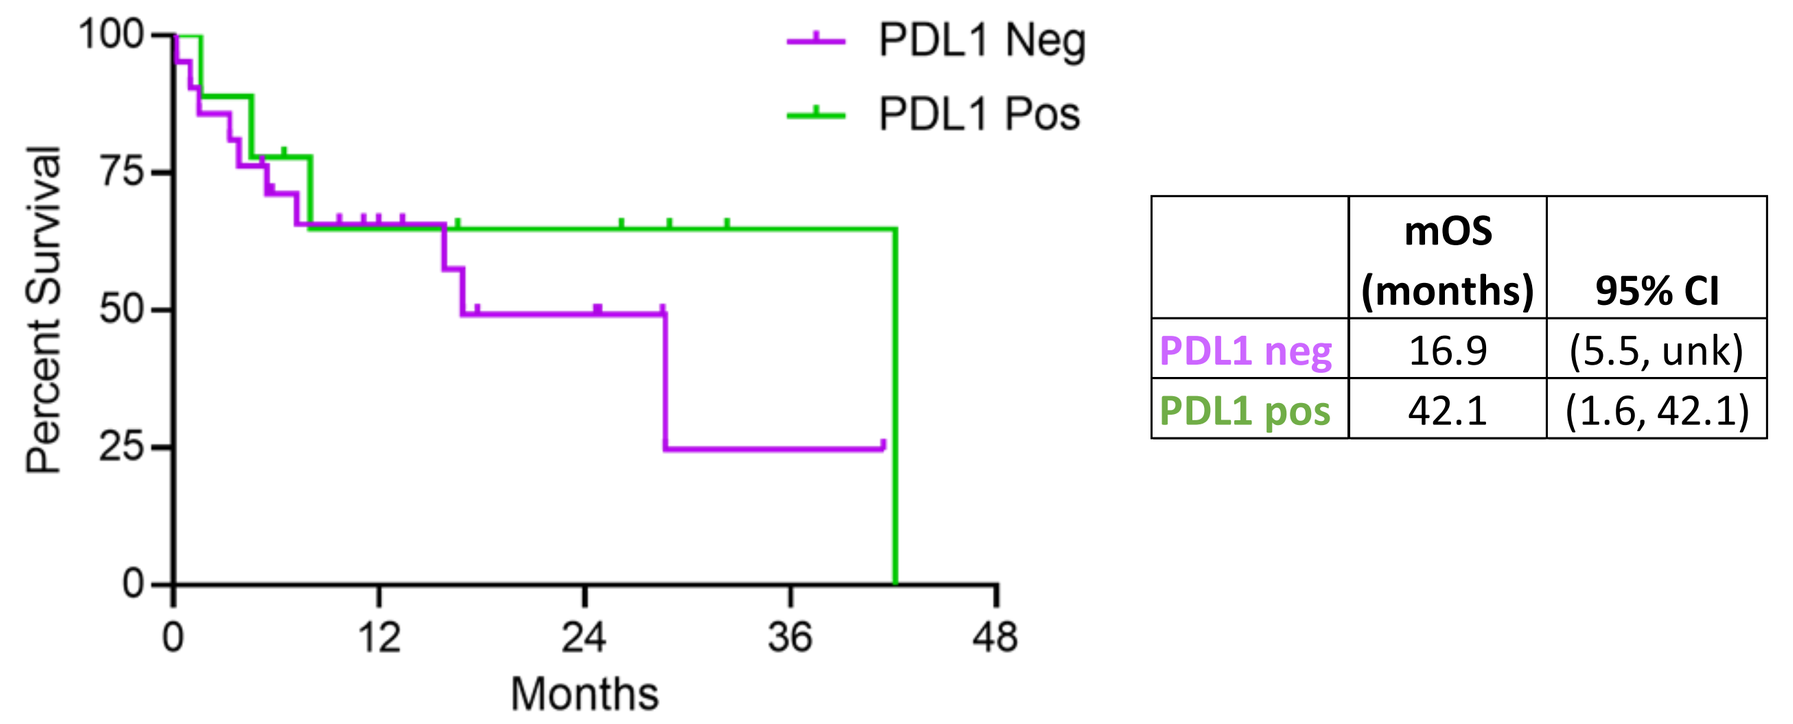
PDL1 Neg
PDL1 Pos
Percent Survival
Months
mOS (months) | 95% CI
PDL1 neg | 16.9 | (5.5, unk)
PDL1 pos | 42.1 | (1.6, 42.1)

Figure 5: Kaplan Meier of estimated Overall Survival – by PDL1 Status
Using a cut-off of 1% PDL1 expression, estimated overall survival is shown for PDL1 negative (n=21) and PDL1 positive (n=9).

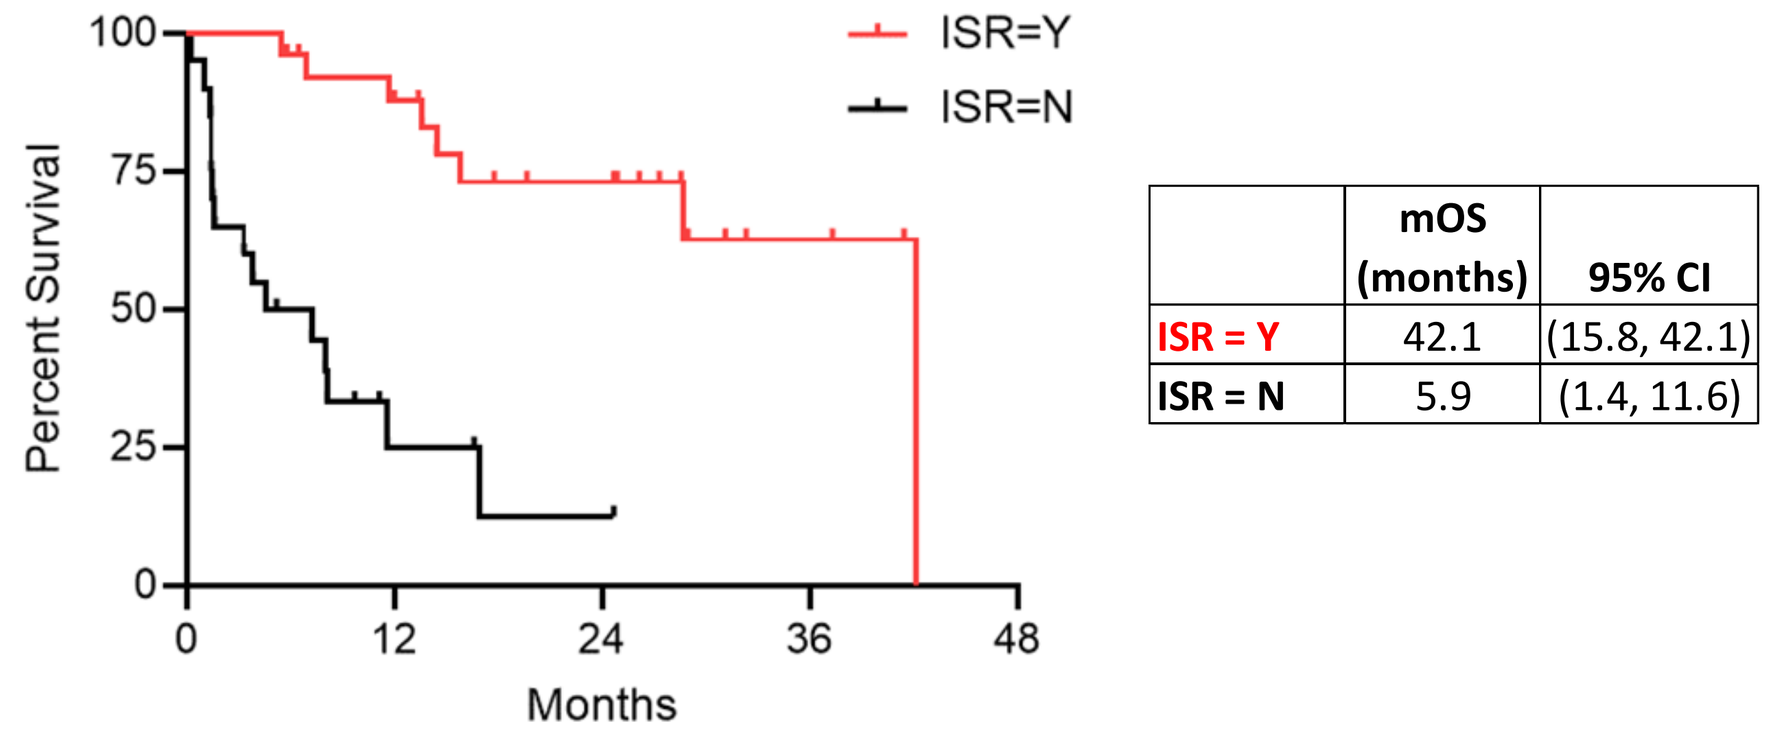
ISR=Y
ISR=N
Percent Survival
Months
mOS (months) | 95% CI
ISR = Y | 42.1 | (15.8, 42.1)
ISR = N | 5.9 | (1.4, 11.6)

Figure 6: Kaplan Meier of estimated Overall Survival – by Injection Site Reaction
Using KM estimates of survival, patients who experienced at least one injection site reaction at any time during treatment (n=26) had statistically significant improved overall survival compared to patients who did not experience an injection site reaction (n=20). HR 0.14 [95% CI; 0.05, 0.36] *p<0.0001*

Best Target Lesion Response

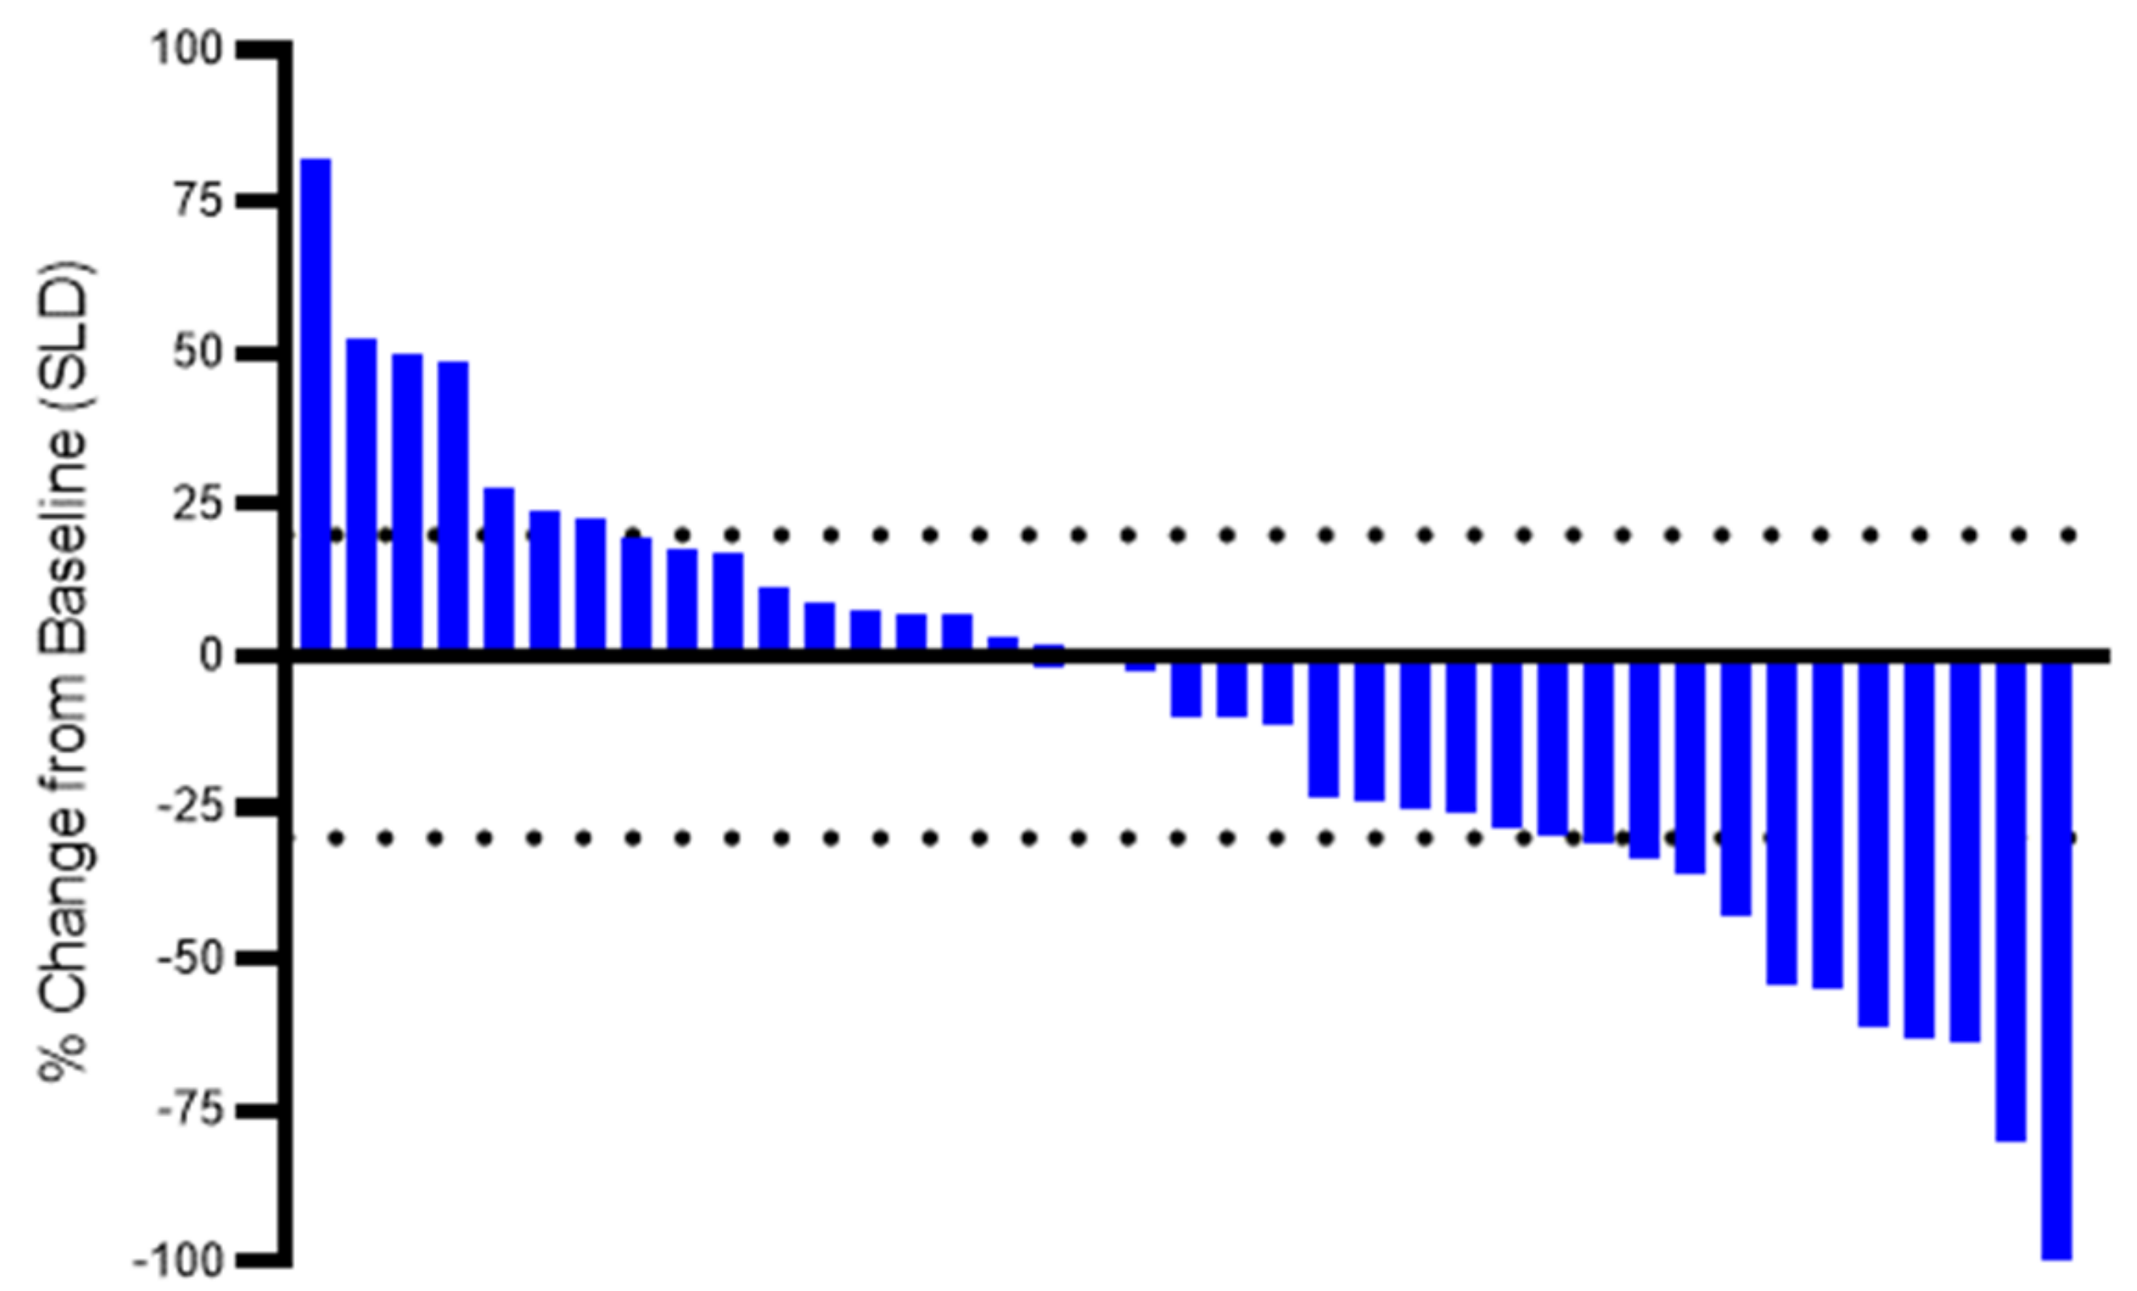
% Change from Baseline (SLD)

Figure 7: Best Target Lesion Response
Waterfall plot of evaluable ITT patients (N=39) using RECIST 1.1 Target Lesion Response. Post-baseline scans not available for 7 patients. Tumor shrinkage was observed in 21 (46%) of 46 ITT patients.

Progression Free Survival

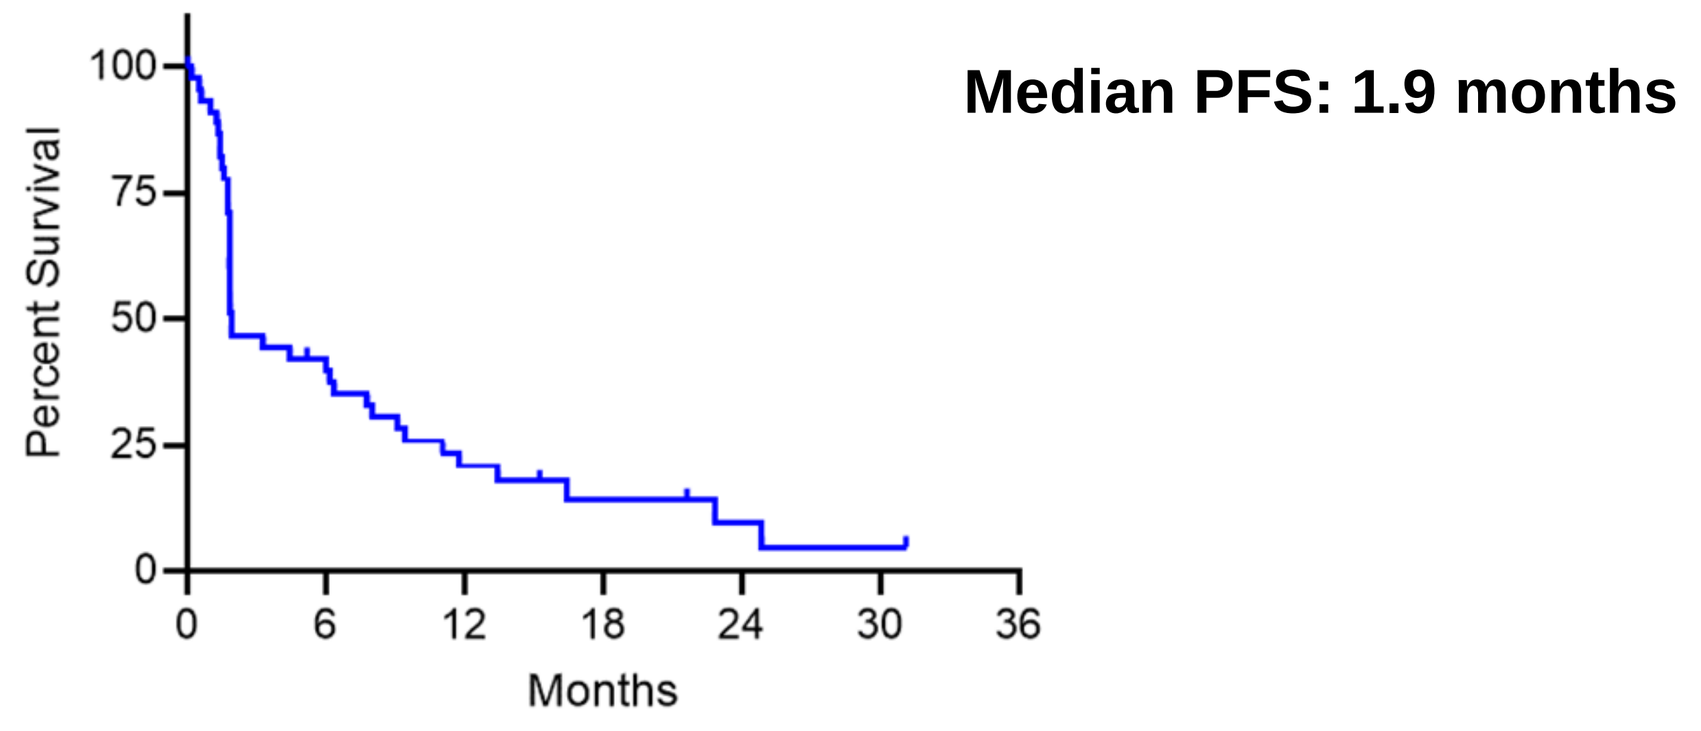
Median PFS: 1.9 months
Percent Survival
Months

Figure 8: Kaplan Meier of Progression-Free Survival – ITT Population
Progression-free survival of ITT population (N=46). Seven (7) patients censored. mPFS is estimated by KM to be 1.9 months [95% CI; 1.8, 6.4].

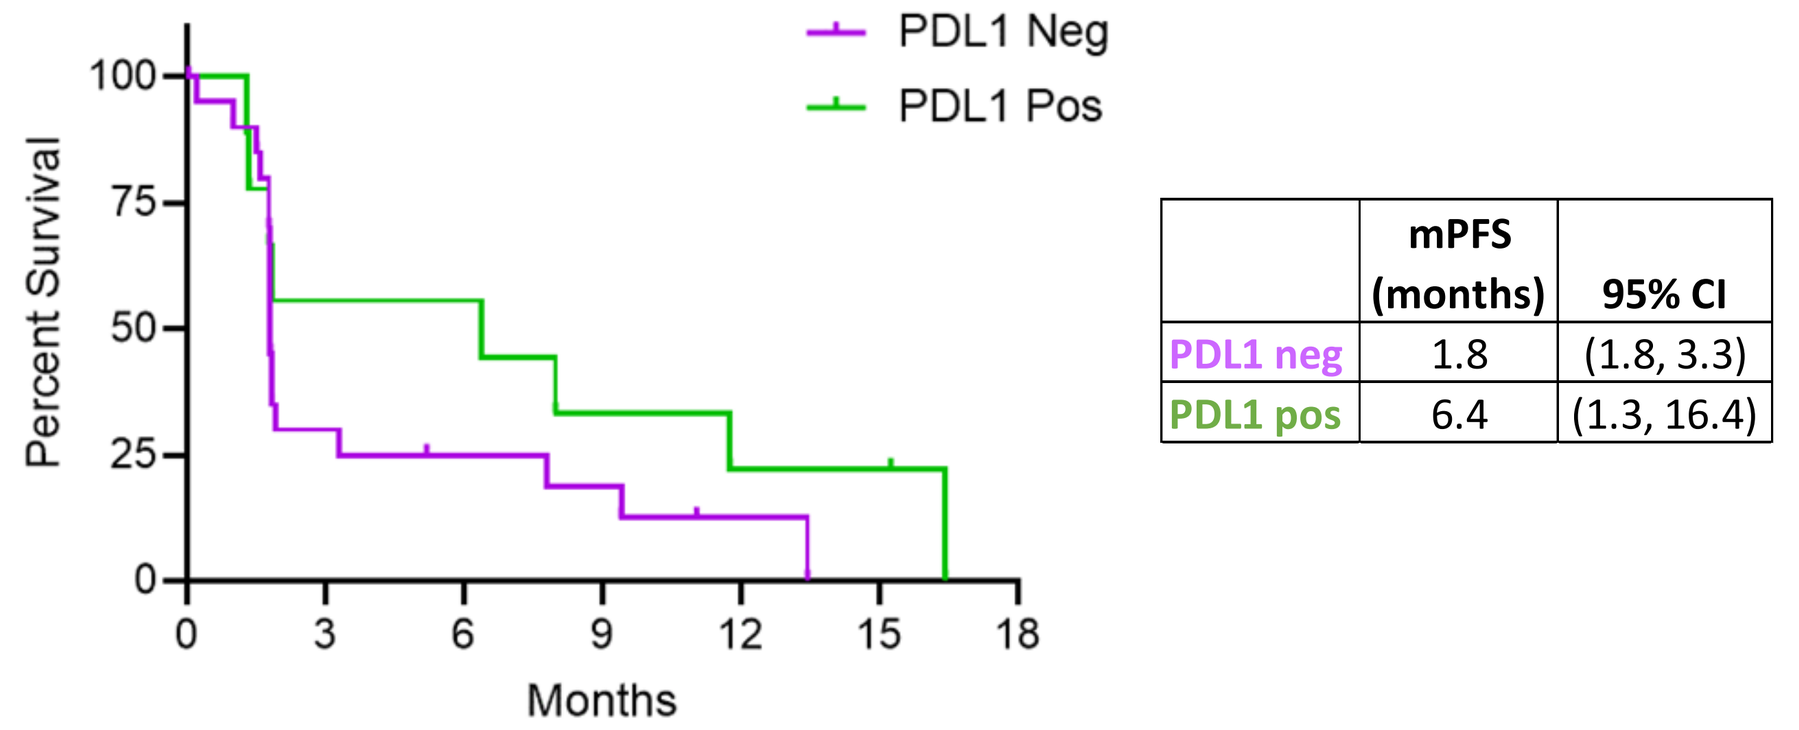
PDL1 Neg
PDL1 Pos
Percent Survival
Months
mPFS (months) | 95% CI
PDL1 neg | 1.8 | (1.8, 3.3)
PDL1 pos | 6.4 | (1.3, 16.4)

Figure 9: Kaplan Meier of Progression-Free Survival – by PDL1 Status
Using a cut-off of 1% PDL1 expression, estimated progression-free survival is shown for PDL1 negative (n=21) and PDL1 positive (n=9).

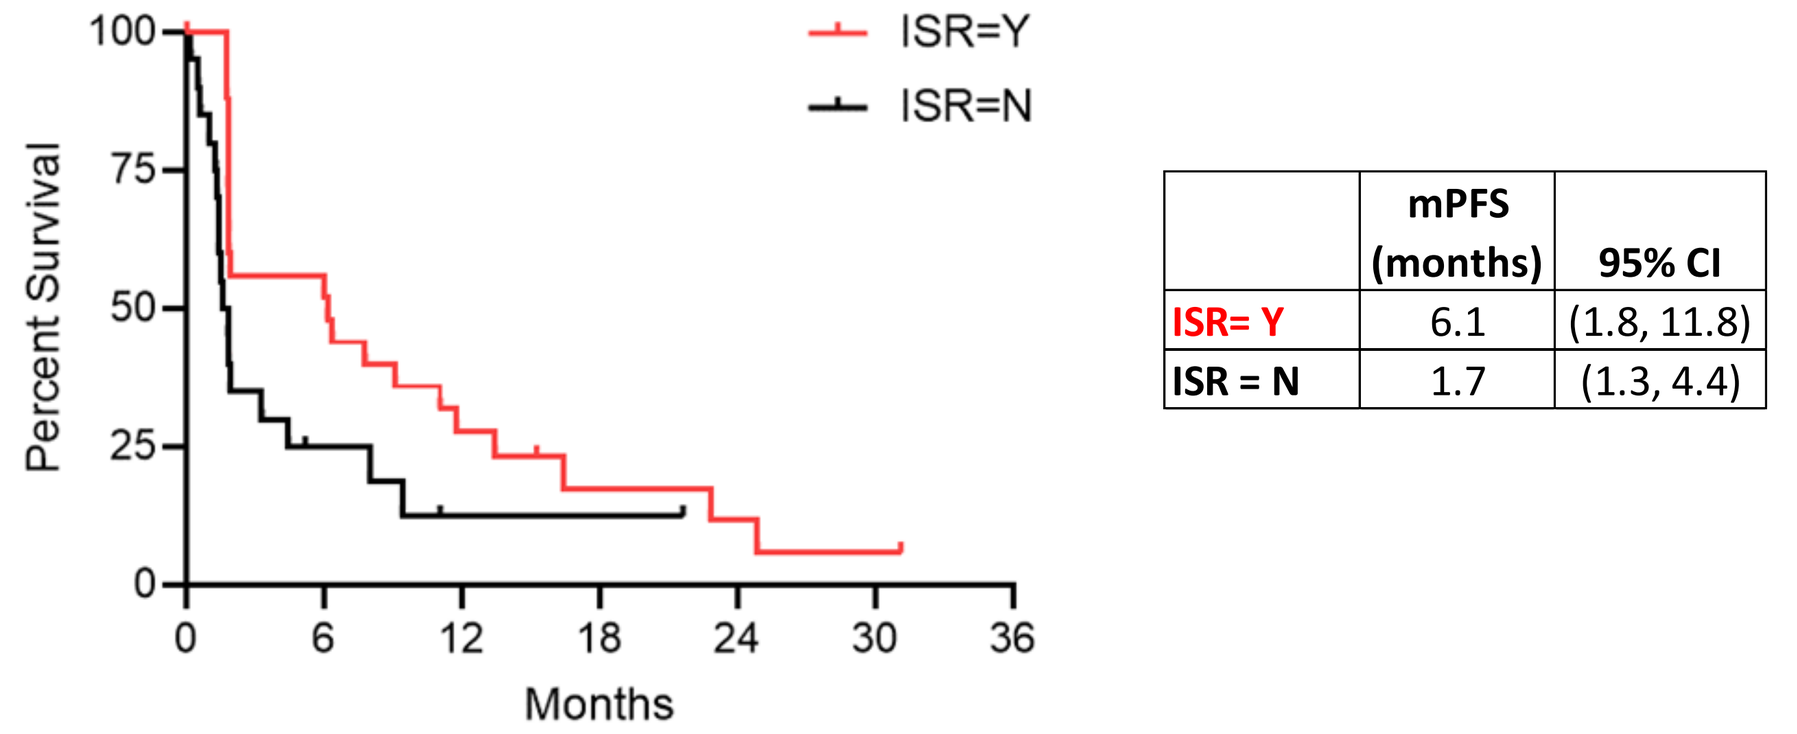
ISR=Y
ISR=N
Percent Survival
Months
mPFS (months) | 95% CI
ISR= Y | 6.1 | (1.8, 11.8)
ISR = N | 1.7 | (1.3, 4.4)

Figure 10: Kaplan Meier of Progression-free Survival – by Injection Site Reaction
Using KM estimates of survival, patients who experienced at least one injection site reaction at any time during treatment (n=26) had statistically significant improved progression-free survival compared to patients who did not experience an injection site reaction (n=20). HR 0.51 [95% CI; 0.26, 0.97] *p=0.0417*

Duration of Clinical Benefit

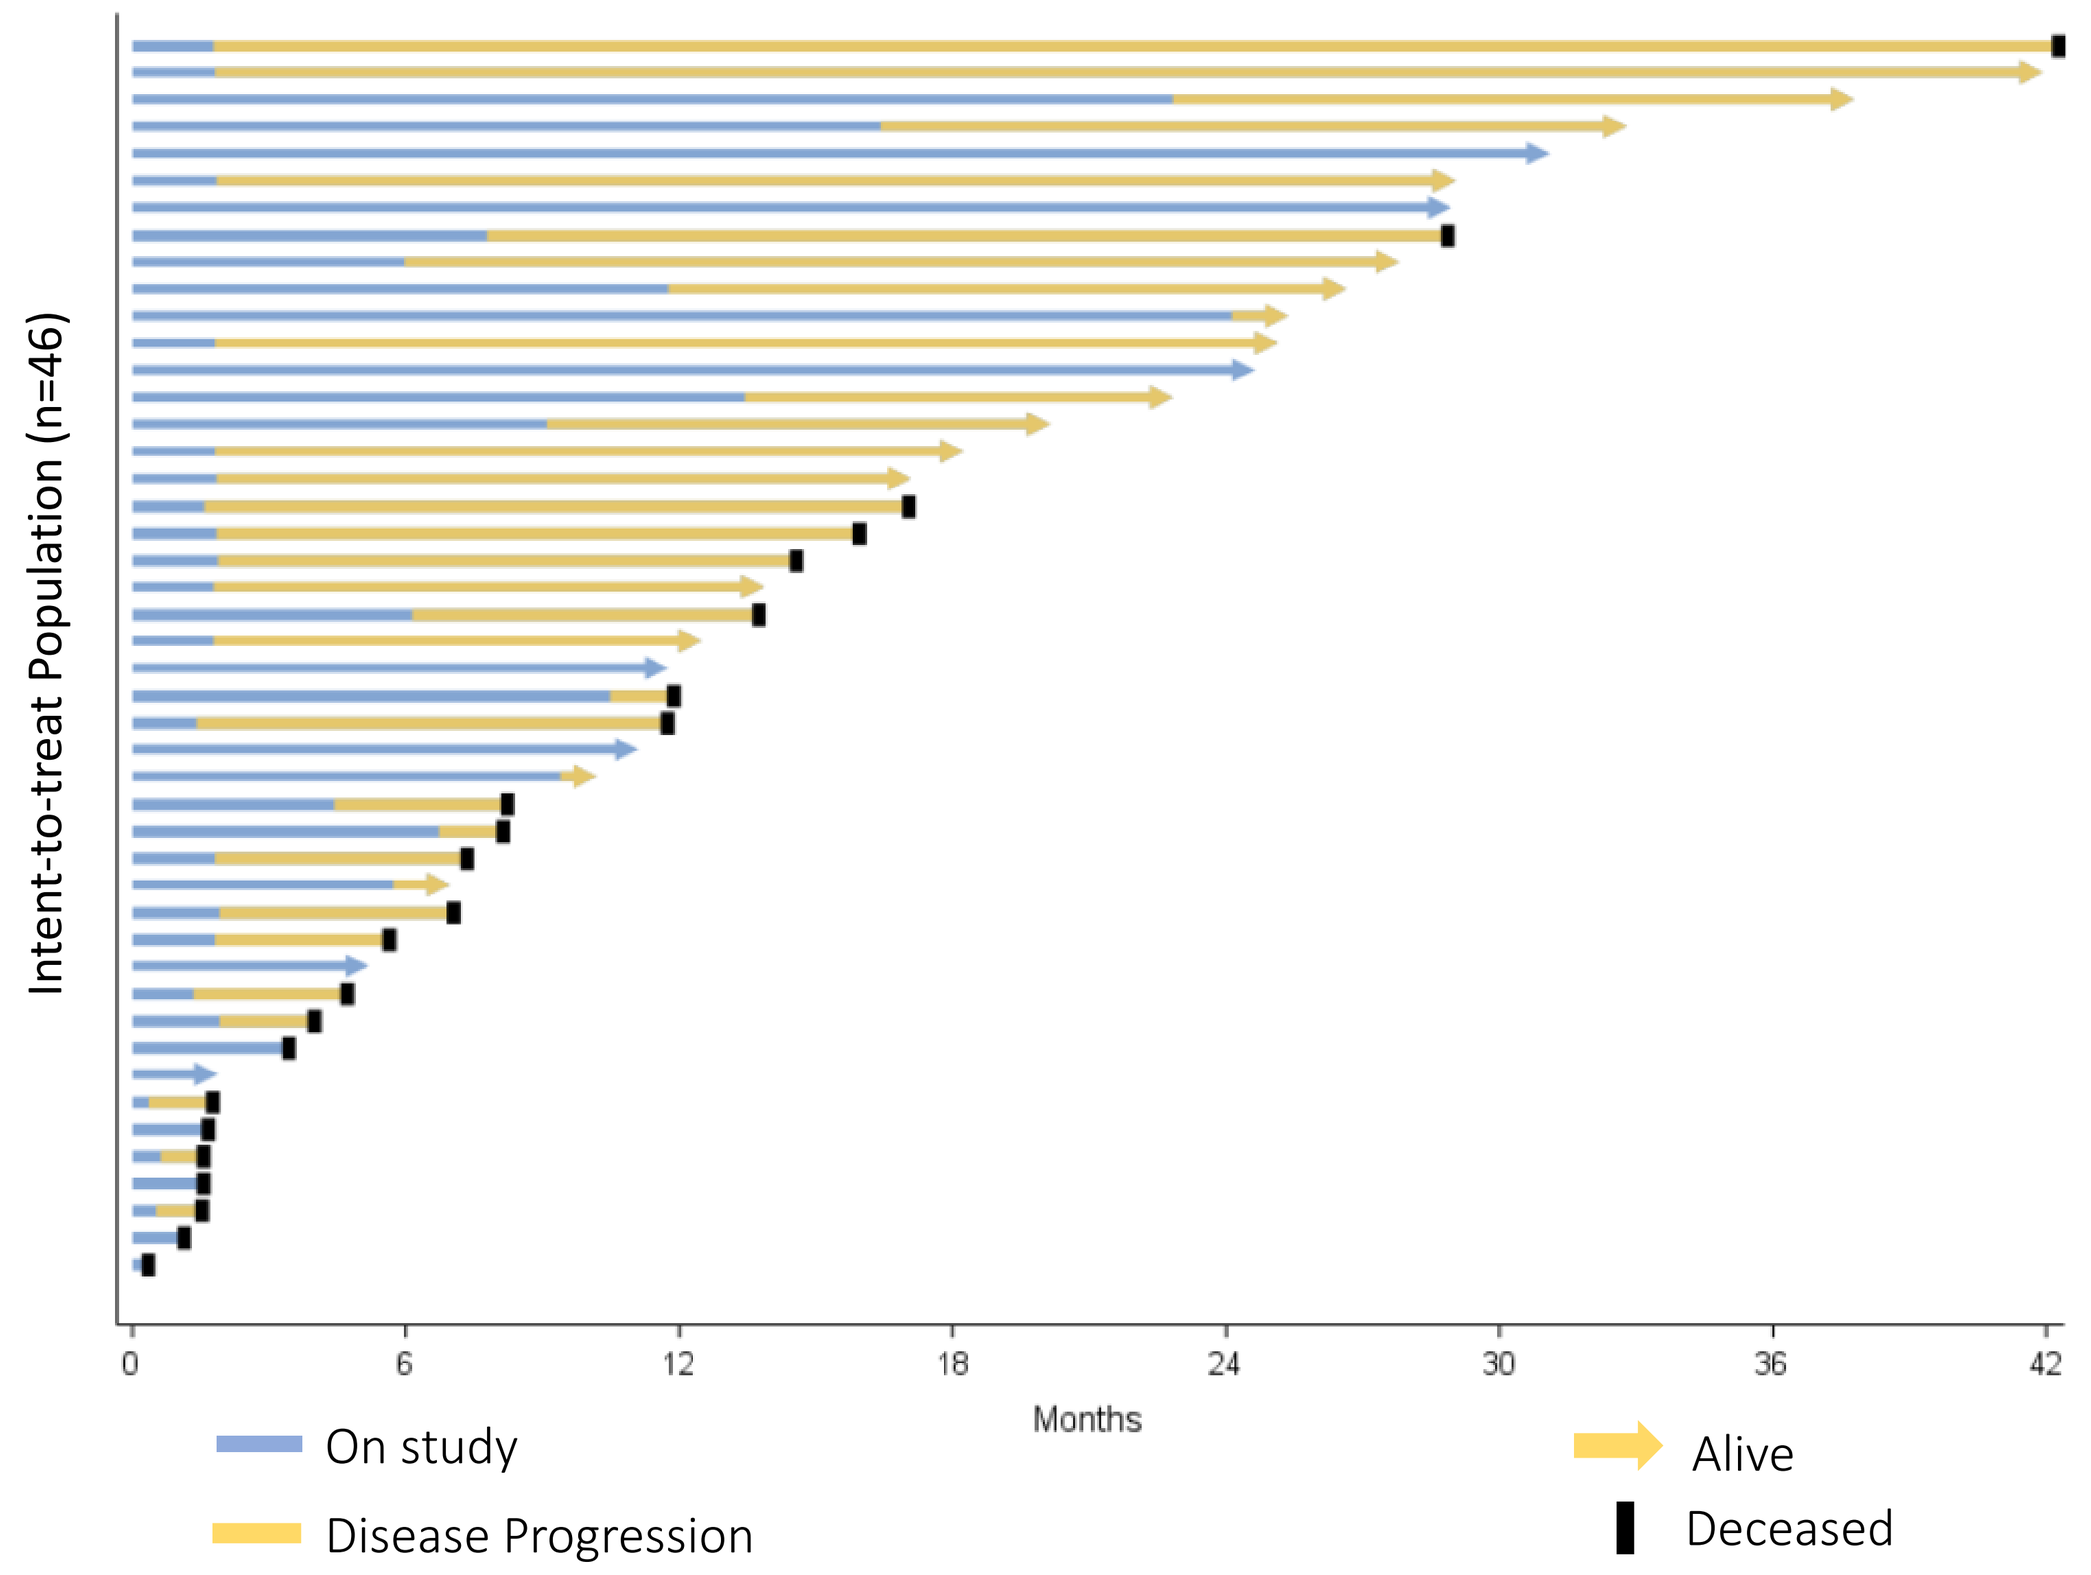
Intent-to-treat Population (n=46)
Months
On study
Disease Progression
Alive
Deceased

Figure 11: Duration of Clinical Benefit
Swimmer plot of time until disease progression and current survival status. With a median follow up time of 17 months, 23 (50%) patients remain alive and 7 (15%) did not experience disease progression.

Frequently Reported Adverse Events

Adverse Events	Cohort A (N=46)
Any Adverse Event	46 (100%)
Any event ≥ Grade 3	17 (37%)
Injection Site Reaction	26 (57%)
Fatigue	12 (26%)
Cough	8 (17%)
Arthralgia	8 (17%)
Constipation	7 (15%)
Diarrhea	7 (15%)
Decreased Appetite	7 (15%)

Table 3: Adverse Event Table
Most commonly reported treatment-emergent adverse events (regardless of attribution) occurring in the safety population. 63% of all AEs were Grade 1 or 2. There was one grade 4 event (hyponatremia) and two grade 5 events (Acute myocardial infarction and Pulmonary embolism due to disease progression) none of which were deemed related to study treatment.

Conclusions

HS-110 in combination with nivolumab is well tolerated.

The effect of HS-110 in combination with nivolumab is not dependent on baseline PDL1 expression.

Best Overall Response of SD or better is associated with on-treatment decreasing levels of terminally differentiated T cell subsets by flow cytometry.

The occurrence of dermal injection site reactions (any grade) is associated with improved progression-free survival and overall survival.

References

1. Strbo N, Garcia-Soto A, Schreiber TH, Podack ER. Secreted heat shock protein gp96-Ig: next-generation vaccines for cancer and infectious diseases. Immunologic research 2013;57:311-25.
2. Oizumi S, Strbo N, Pahwa S, Deyev V, and Podack ER. Molecular and cellular requirements for enhanced antigen cross-presentation to CD8 cytotoxic T lymphocytes. Journal of immunology 2007; 179, 2310-2317.

Acknowledgements

The authors would like to acknowledge the following Heat employees for their efforts and contributions in producing the translational clinical data in support of this clinical trial: Elena Gorovits, Beata Boczkowska, David Taylor and Matthew Seavey.

The authors are grateful for the Investigators, study staff, patients and their families for the commitment to this trial to help advance the treatment of non-small cell lung cancer.

For study-related correspondence, contact lmcdermott@heatbio.com