A Phase 1a/b Dose Escalation Study of the Mutation Agnostic FLT3/BTK Inhibitor Luxeptinib (CG-806) in Patients with Relapsed or Refractory Acute Myeloid Leukemia

Aaron D. Goldberg[1], Maro Ohanian[2], Paul B. Koller[3], Jessica Altman[4], Mohamad Cherry[5], Benjamin Tomlinson[6], Namrata Chandhok[7], Hongying Zhang[8], Nasrin Rastgoo[8], Khalid Benbatoul[8], Yuying Jin[8], Genia Su[8], Donna N. Haney[8], Jotin Marango[8], Stephen Howell[9], William Rice[8], Rafael Bejar[8,9]

[1]Department of Medicine, Leukemia Service, Memorial Sloan-Kettering Cancer Center, New York, NY; [2]Department of Leukemia, The University of Texas MD Anderson Cancer Center, Houston, TX; [3] Division of Leukemia, Department of Hematology and HCT, City of Hope National Medical Center, Duarte, CA; [4]Robert H. Lurie Comprehensive Cancer Center of Northwestern University, Chicago, IL; [5]Morristown Medical Center, Morristown, NJ; [6]University Hospital of Cleveland, Cleveland, OH; [7]Sylvester Comprehensive Cancer Center, University of Miami, Miami, FL; [8]Aptose Biosciences Inc, San Diego, CA; [9]Moores Cancer Center, University of California San Diego Health, San Diego, CA

INTRODUCTION

Luxeptinib (CG-806) is a potent oral small molecule inhibitor of the wild type and all mutant forms of the FLT3 kinase, including ITD, D835Y, and F691L. Luxeptinib simultaneously suppresses additional signaling pathways in AML cells (CSF1R, PDGFRα, TRK, SYK, BTK, LYN, AKT, ERK, MAPK), kills primary AML cells insensitive to other FLT3 inhibitors at pM and low nM concentrations, and shows enhanced activity in combination with venetoclax. Patient-derived AML cells retain sensitivity to luxeptinib even when harboring mutations of *NPM1*, *IDH1*, *ASXL1*, or *TP53*

Selectively & Potently Inhibits Myeloid & Lymphoid Kinomes

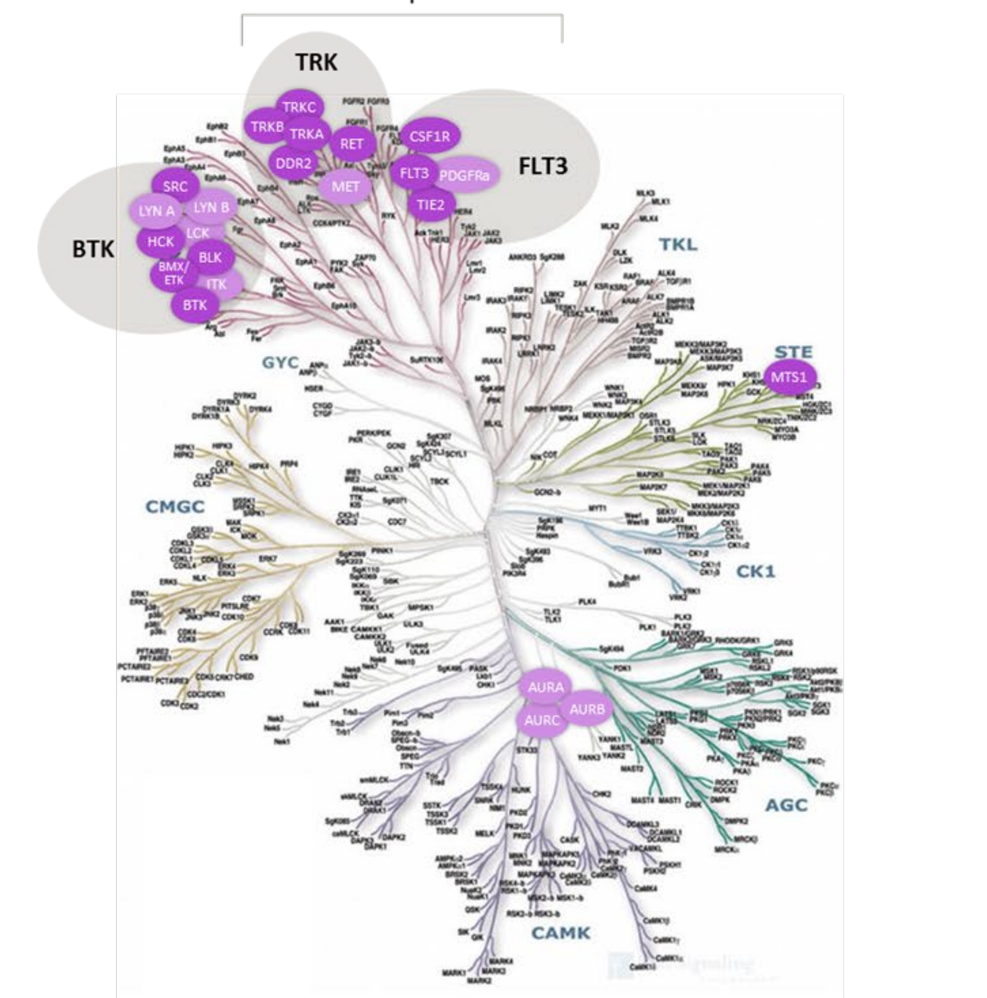
Receptors
TRK
FLT3
BTK
TKL
STE
GYC
CMGC
CK1
AGC
CAMK

Robust Preclinical Efficacy
Covered Key Mutational Subgroups in AML*

FLT3	Lux potently inhibits both the wild type and all mutant forms of FLT3
NPM1	Lux inhibits **SYK** phosphorylation and efficiently suppresses its downstream pathways
IDH1	AML patient samples with IDH1 mutations are more sensitive to Lux than IDH WT
TP53	AML patient samples with wild type and mutant TP53 remain sensitive to Lux
NRAS	AML patient samples with wild type and mutant NRAS remain sensitive to Lux
ASXL1	AML patient samples with wild type and mutant ASXL1 are equally sensitive to Lux
KIT	Lux potently inhibits both the wild type and certain mutant forms of KIT

Patient Demographics

Patient Demographics	Cohorts 1 to 3 (N=16)*
Median Age (Range), Years	**73.5 (36, 81)**
Sex, N (%)	
Male	**11 (68.8%)**
Female	**5 (31.3%)**
Ethnicity, N (%)	
Not Hispanic or Latino	**12 (75.0%)**
Hispanic or Latino	**1 (6.3%)**
Not Reported	**3 (18.8%)**
Race, N (%)	
White	**12 (75.0%)**
Asian	**2 (12.5%)**
Native Hawaiian or Other Pacific Islander	**1 (6.3%)**
Other	**1 (6.3%)**
ECOG Score, N (%)	
0 -Normal activity	**2 (12.5%)**
1 -Symptoms, but ambulatory	**13 (81.3%)**
2 -In bed <50% of the time	**1 (6.3%)**
FLT3 Mutation Status, N (%)	
WT	**3 (18.8%)**
ITD	**11 (68.8%)**
TKD or gatekeeper mutations	**4 (25.0%)**
AML Type, N (%)	
De novo	**11 (68.8%)**
Secondary AML	**4 (25.0%)**
Therapy-related AML	**1 (6.3%)**
Relapsed or Refractory, N (%)	
Relapsed	**3 (18.8%)**
Refractory	**5 (31.3%)**
Both Relapsed and Refractory	**8 (50.0%)**
RBC Transfusion Dependent, N (%)	
Yes	**12 (75.0%)**
Platelet Transfusion Dependent, N (%)	
Yes	**11 (68.8%)**
Median Number of Lines of Prior Therapy (Range)	**3 (1, 8)**
Chemotherapy, N(%)	**10 (62.5%)**
Transplant, N (%)	**3 (18.8%)**
Radiation, N(%)	**1 (6.3%)**
Targeted and Immunotherapy, N (%)	
Hypomethylating Agent	**16 (100%)**
Anti-BCL2 (venetoclax)	**15 (93.8%)**
FLT3 Inhibitor**	**10 (62.5%)**
Antibody drug conjugate	**2 (12.5%)**
IDH1-Inhibitor (ivosidenib)	**1 (6.3%)**
JAK Inhibitor (ruxolitinib)	**1 (6.3%)**
Checkpoint Inhibitor (ipilimumab)	**1 (6.3%)**
Other Experimental Agent	**2 (12.5%)**

*Data-cut date: Oct 06, 2021
**9 patients recieved gilteritinib, 5 of them also received other FLT3i crenolanib, quizartinib, or midostaurin. 1 patient received sorafenib.

OBJECTIVES & STUDY DESIGN

(Ongoing Phase 1 a/b, open-label, single arm, multicenter, 3+3 dose-escalation clinical study (NCT04477291) in **Relapsed or refractory AML and higher-risk MDS** who **failed or are ineligible for / intolerant of** intensive chemotherapy or transplantation

- Patients administered **oral capsules, twice daily** on a **28-day cycle**
- Planned expansion cohorts after dose escalation
- Additional patients may be enrolled (**back filling**) at dose levels previously declared safe
- **Intra-patient dose escalation** is allowed if higher dose is safe in 3 or more patients

Primary objectives:

- Assess safety and tolerability of luxeptinib (CG-806)
- Determine maximum tolerated dose (MTD) and / or recommended Phase 2 dose (RP2D)

Key secondary objectives:

- Assess PK profile and PD activity
- Obtain preliminary evidence of antitumor activity

Cohort	Dose	Status
1	450 mg BID	Completed
2	600 mg BID	Completed
3	750 mg BID	Completed
4	**900 mg BID**	**Ongoing**
5	1050 mg BID	Planned
6	1200 mg BID*	Planned

*Protocol enables dose escalation directly from Cohort 900mg to Cohort 1200mg based upon decision of CSRC.

Safety and Tolerability Profile

- **Luxeptinib was generally well tolerated across 450 – 750mg BID over multiple cycles.**
- **Currently, dose level 900mg BID (Cohort 4) is ongoing.**
- **One DLT occurred in each dose level of 450mg and 750mg (scored as "possibly related" to study drug), requiring expansion to 6 patients. No DLTs occurred in the other 5 patients and supported dose escalation.**

Events	Cohorts 1 to 3 (N=16)*
Any Treatment Emergent Adverse Events (TEAEs)	15 (93.8%)
Any TEAEs ≥ Grade 3	14 (87.5%)
Any Luxeptinib **Related** TEAEs ≥ Grade 3	4 (25.0%)
TEAE Leading to Treatment Discontinuation	3 (18.8%)
TEAE Leading to Death	3 (18.8%)
Any Luxeptinib **Related** TEAEs Leading to Death	0 (0%)
Any Serious TEAEs (SAEs)	13 (81.3%)
Any Luxeptinib **Related** SAEs	2 (12.5%)†
Dose Limiting Toxicity	2 (12.5%)††

* Data-cut date: Oct 06, 2021
† One patient had Grade 3 encephalopathy; another patient had Grade 3 pericardial effusion and Grade 2 pleural effusion. All were assessed as **possibly** related to Lux.
†† The above mentioned two Grade 3 SAEs in two patients were assessed as DLTs.

Luxeptinib Related Treatment Emergent Adverse Events

Preferred Term	Cohorts 1 to 3 (N=16)*		
	Any Grade, N (%)	Grade 3, N (%)	Grade 4, N (%)
Any Related TEAE	9 (56.3%)	2 (12.5%)	2 (12.5%)
Fatigue	3 (18.8%)	0	0
Nausea	3 (18.8%)	0	0
Anaemia	2 (12.5%)	2 (12.5%)	0
Blood alkaline phosphatase increased	2 (12.5%)	0	0
Platelet count decreased	2 (12.5%)	0	2 (12.5%)
Abdominal distension	1 (6.3%)	0	0
Abdominal pain	1 (6.3%)	0	0
Activated partial thromboplastin time prolonged	1 (6.3%)	0	0
Constipation	1 (6.3%)	0	0
Decreased appetite	1 (6.3%)	0	0
Diarrhoea	1 (6.3%)	0	0
Encephalopathy	1 (6.3%)	1 (6.3%)	0
Headache	1 (6.3%)	0	0
Hyperphosphataemia	1 (6.3%)	0	0
Insomnia	1 (6.3%)	0	0
Lymphocyte count decreased	1 (6.3%)	1 (6.3%)	0
Neutrophil count decreased	1 (6.3%)	0	1 (6.3%)
Pericardial effusion	1 (6.3%)	1 (6.3%)	0
Photophobia	1 (6.3%)	0	0
Pleural effusion	1 (6.3%)	0	0
Weight decreased	1 (6.3%)	0	0
White blood cell count decreased	1 (6.3%)	1 (6.3%)	0

*No luxeptinib related TEAEs = Grade 5 as of Oct 06, 2021

ASH2021 Abstract# 1272

Treatment Cohorts

As of data-cut date on October 6, 2021:

- 16 patients were treated across 3 cohorts:
 11 FLT3-ITD (69%), 4 FLT3-TKD/gatekeeper mutant (25%) including 2 ITD+F691L, 1 D835Y, 1 D698H, 3 FLT3 WT
- Heavily-pretreated AML patients with **median 3 lines** of prior therapies (range 1-8)
- **10 (62.5%)** patients had prior FLT3i therapy.
 9 (56%) received gilteritinib, 5 of them also received other FLT3i including midostaurin, quizartinib or crenolanib; 1 patient received sorafenib.
- 4 patients in DDI study co-administered with a CYP3A4/5 inhibiting azole anti-fungal.
- 4 patients continue treatment on study

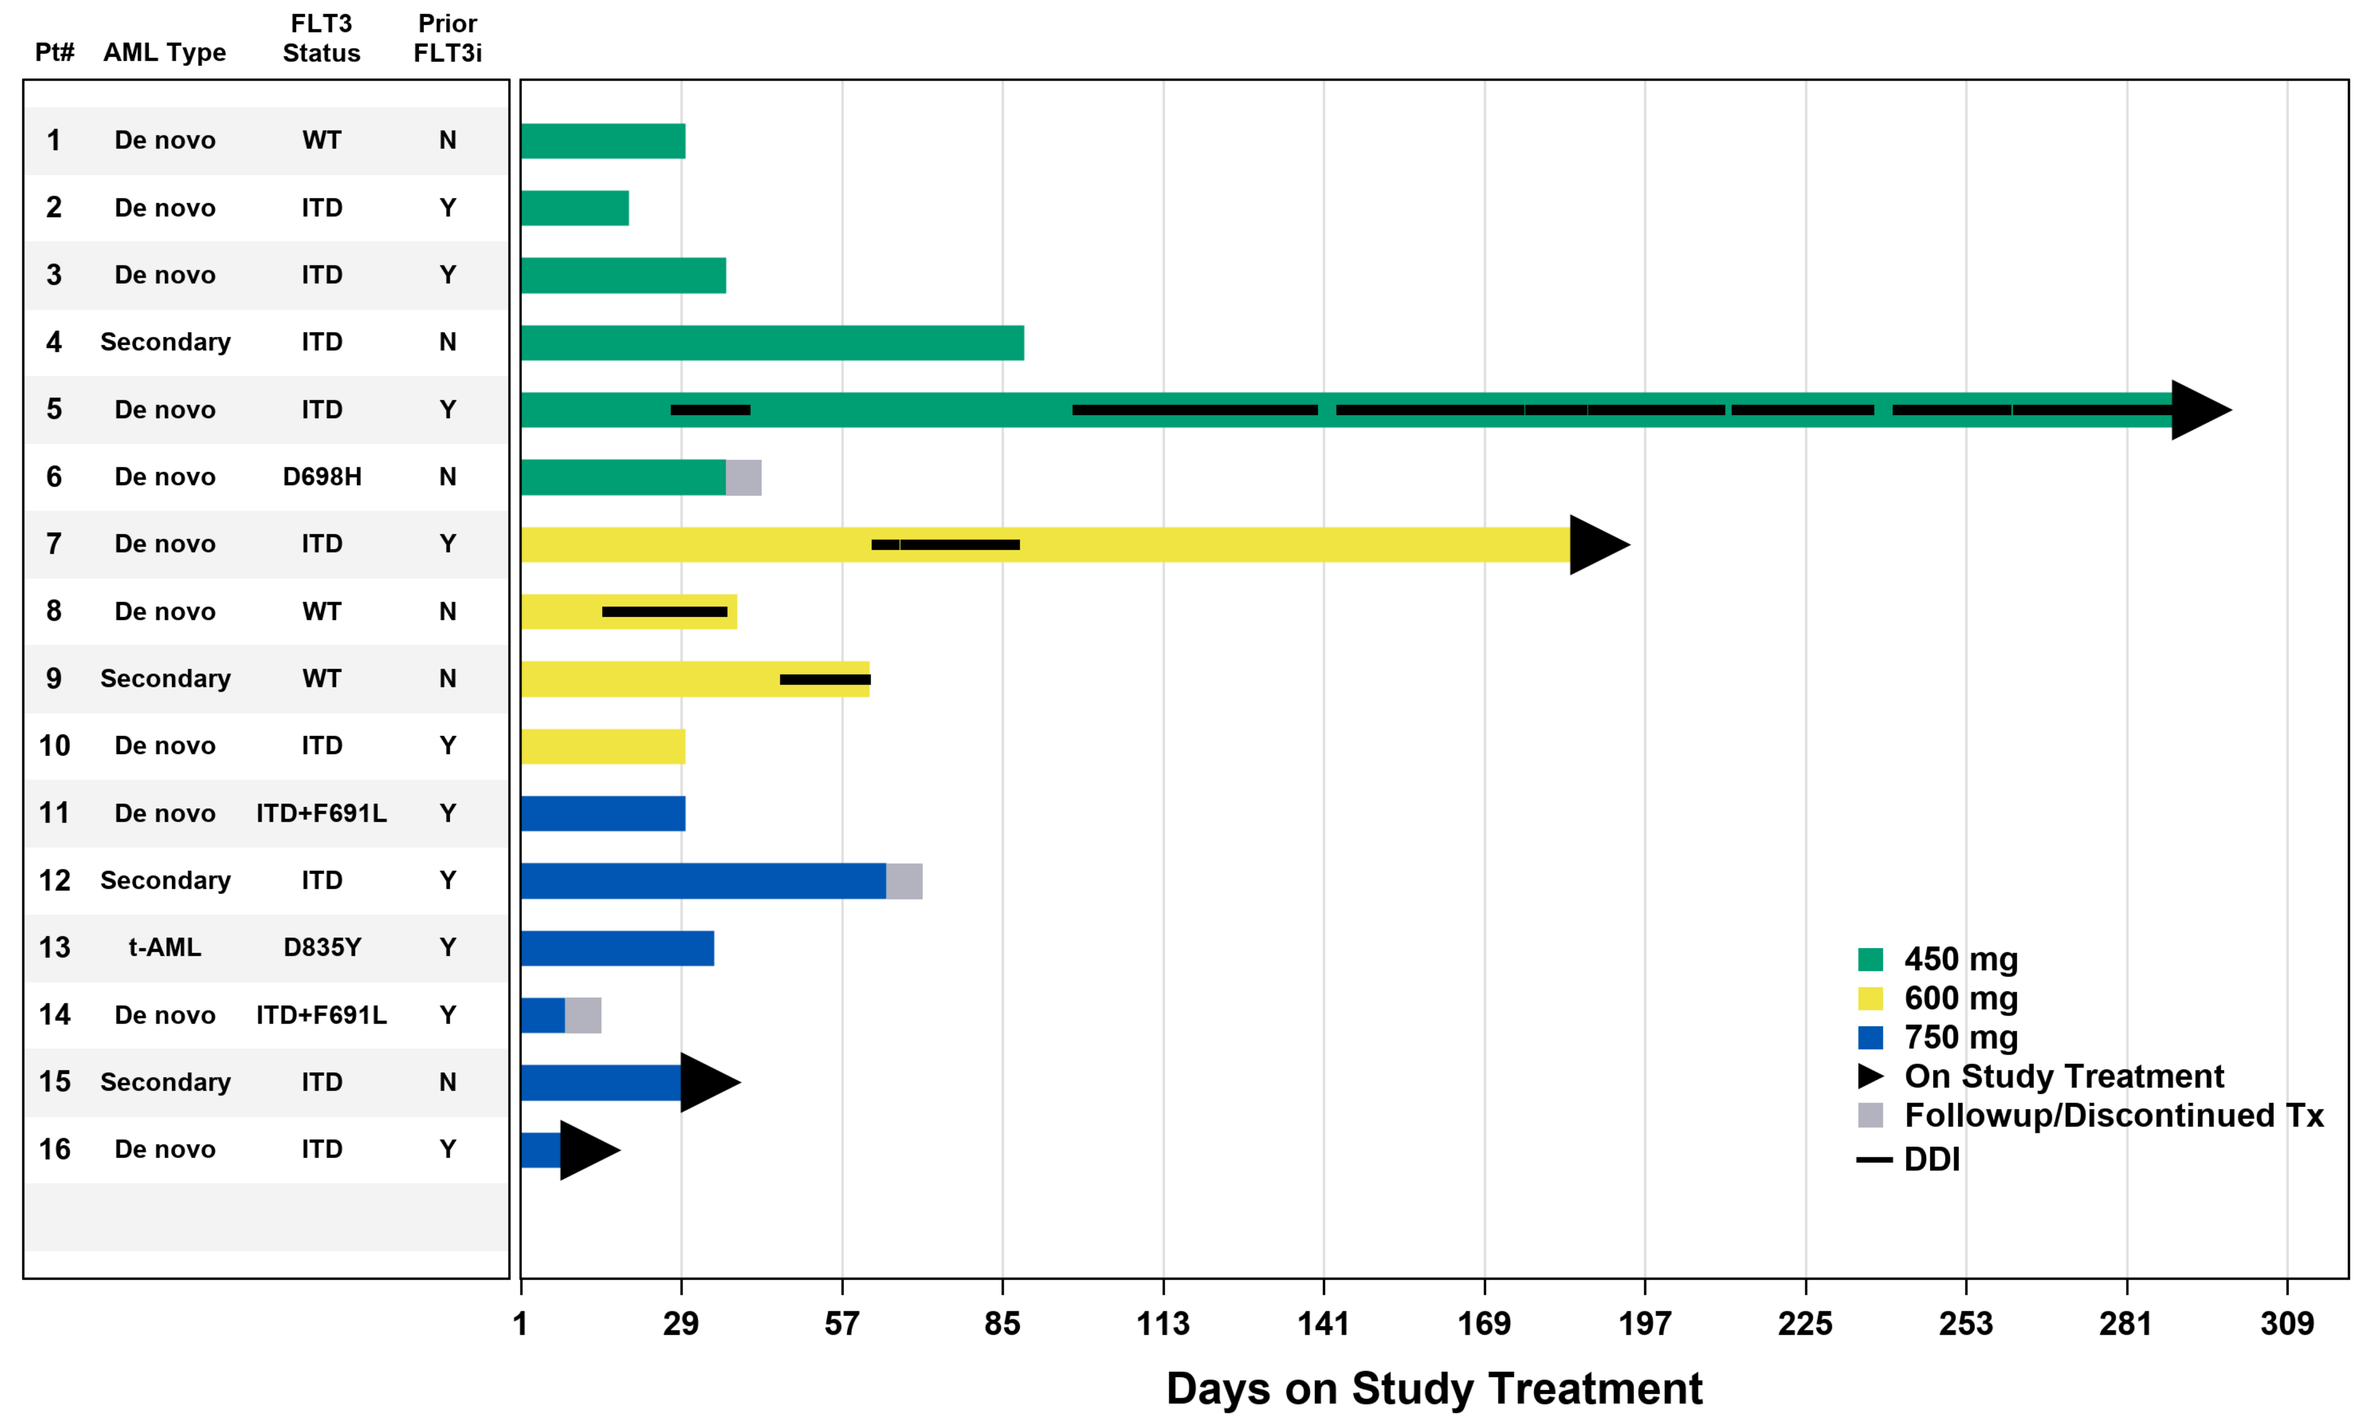
Pt# AML Type FLT3 Status Prior FLT3i
1 De novo WT N
2 De novo ITD Y
3 De novo ITD Y
4 Secondary ITD N
5 De novo ITD Y
6 De novo D698H N
7 De novo ITD Y
8 De novo WT N
9 Secondary WT N
10 De novo ITD Y
11 De novo ITD+F691L Y
12 Secondary ITD Y
13 t-AML D835Y Y
14 De novo ITD+F691L Y
15 Secondary ITD N
16 De novo ITD Y
450 mg
600 mg
750 mg
On Study Treatment
Followup/Discontinued Tx
DDI
1 29 57 85 113 141 169 197 225 253 281 309
Days on Study Treatment

Luxeptinib Pharmacodynamic Biomarker: FLT3 Target Engagement in R/R AML Patients

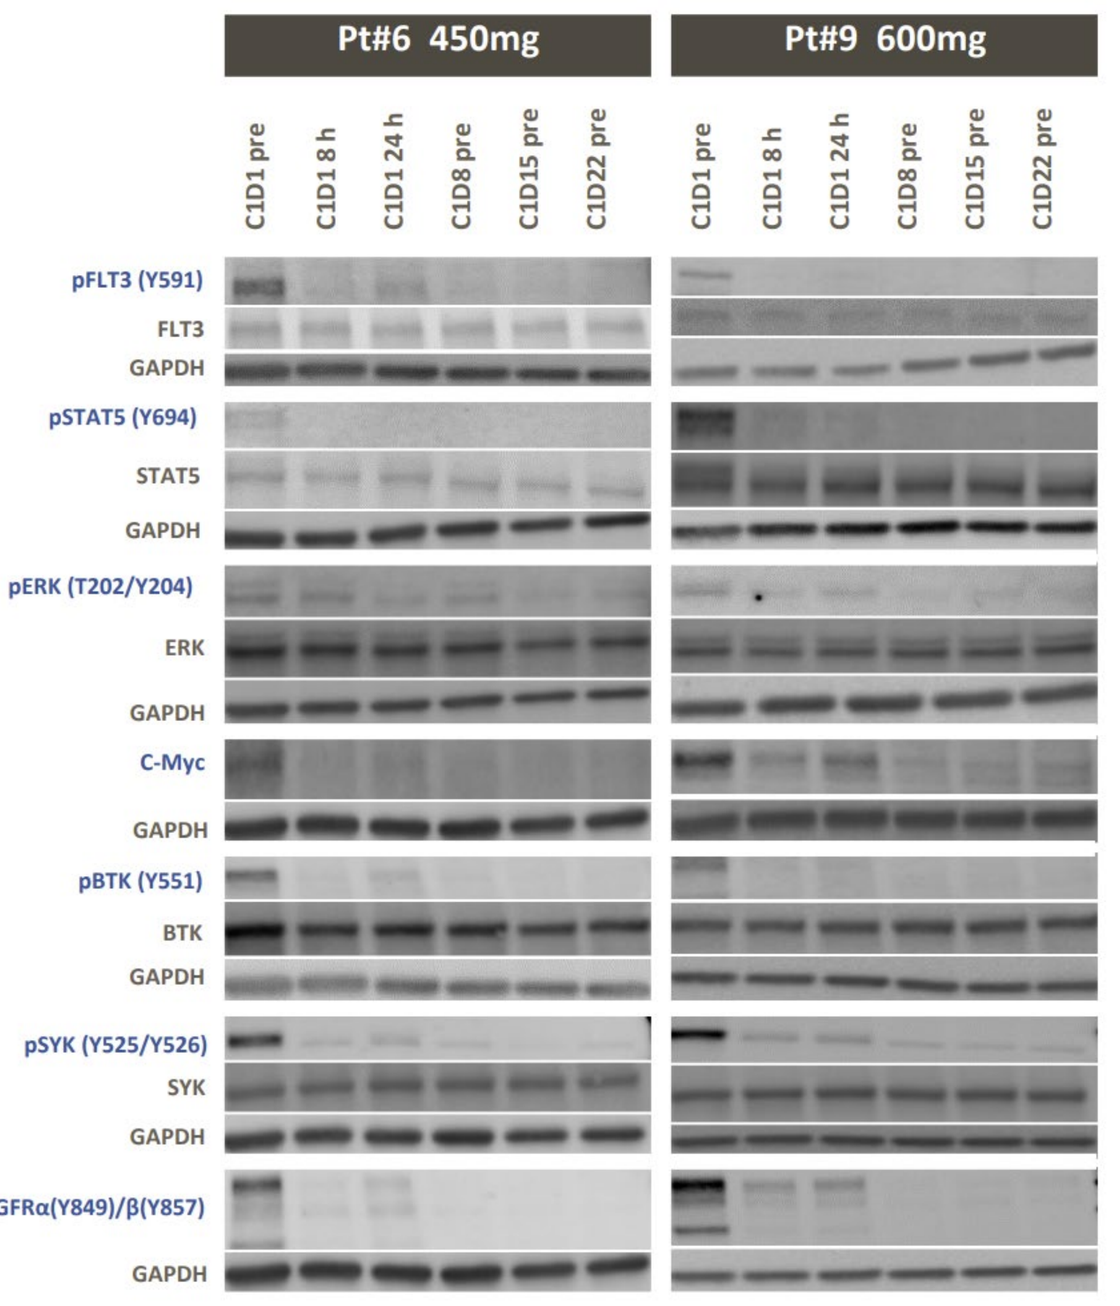
Pt#6 450mg
Pt#9 600mg
C1D1 pre
C1D1 8 h
C1D1 24 h
C1D8 pre
C1D15 pre
C1D22 pre
pFLT3 (Y591)
FLT3
GAPDH
pSTAT5 (Y694)
STAT5
GAPDH
pERK (T202/Y204)
ERK
GAPDH
C-Myc
GAPDH
pBTK (Y551)
BTK
GAPDH
pSYK (Y525/Y526)
SYK
GAPDH
pPDGFRα(Y849)/β(Y857)
GAPDH

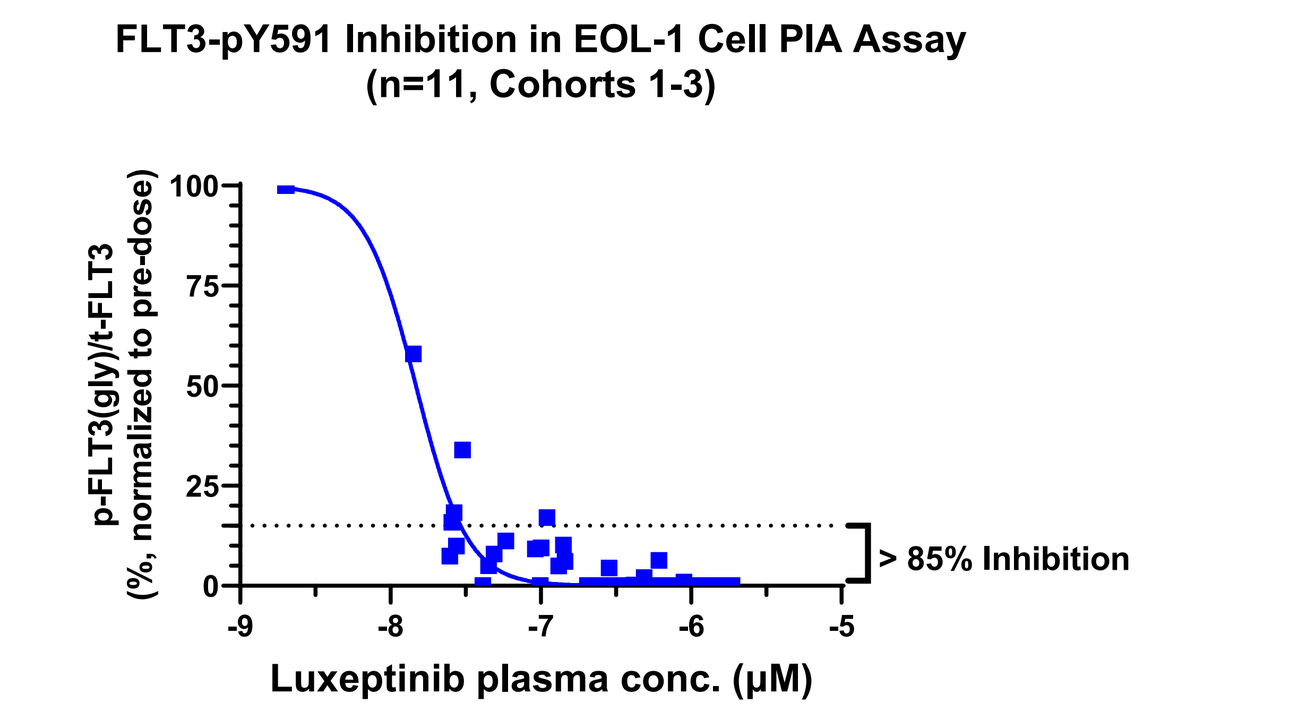
FLT3-pY591 Inhibition in EOL-1 Cell PIA Assay
(n=11, Cohorts 1-3)
p-FLT3(gly)/t-FLT3 (%, normalized to pre-dose)
> 85% Inhibition
Luxeptinib plasma conc. (μM)

FLT3 target engagement by luxeptinib in plasma inhibitory activity (PIA) assay, a surrogate for in vivo FLT3 inhibition

- Plasma levels >0.2 μM consistently deliver > 85% inhibition of FLT3-pY591 in reporter cells EOL-1
- Dose-dependent inhibition of FLT3 signaling (pFLT3, pSTAT5, pERK and c-Myc)
- Dose-dependent inhibition of non-FLT3 pathways phospho- SYK, BTK and PDGFα.

Plasma PK Profile

Luxeptinib achieved plasma concentrations C_{trough} **around 1μM** for dose levels of 600mg (cohort 1) and 750mg (cohort 3)

Cycle 1 Day 1

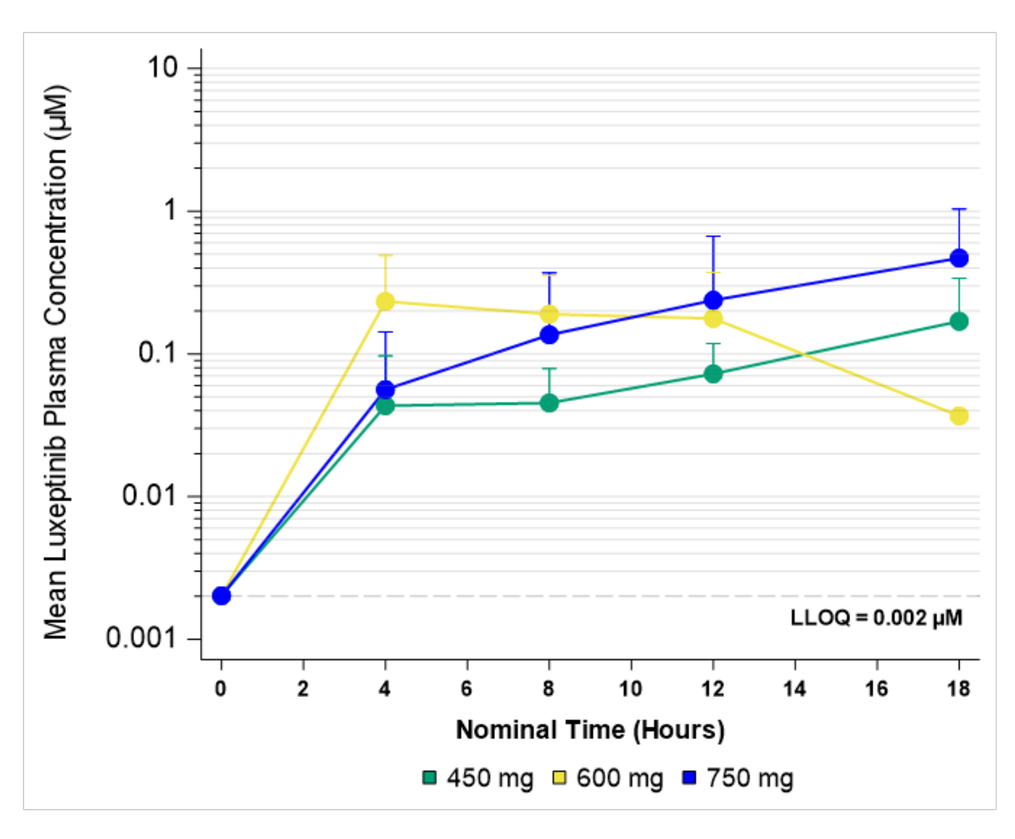
Mean Luxeptinib Plasma Concentration (μM)
LLOQ = 0.002 μM
Nominal Time (Hours)
450 mg 600 mg 750 mg

Cycle 1 (28 Days)

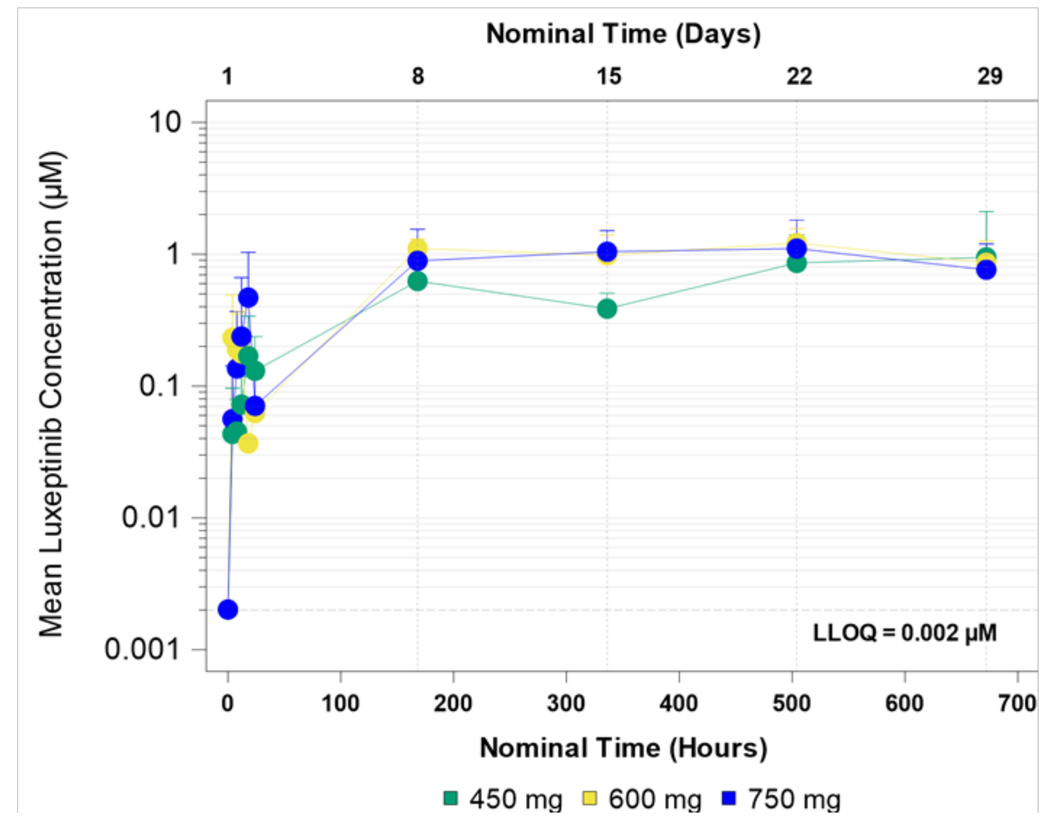
Nominal Time (Days)
Mean Luxeptinib Concentration (μM)
LLOQ = 0.002 μM
Nominal Time (Hours)
450 mg 600 mg 750 mg

We thank our principal investigators, clinical site staff, and most importantly, our patients and their families for their participation in this clinical trial.

Pharmacokinetic Profile

Antitumor Activity in Patients

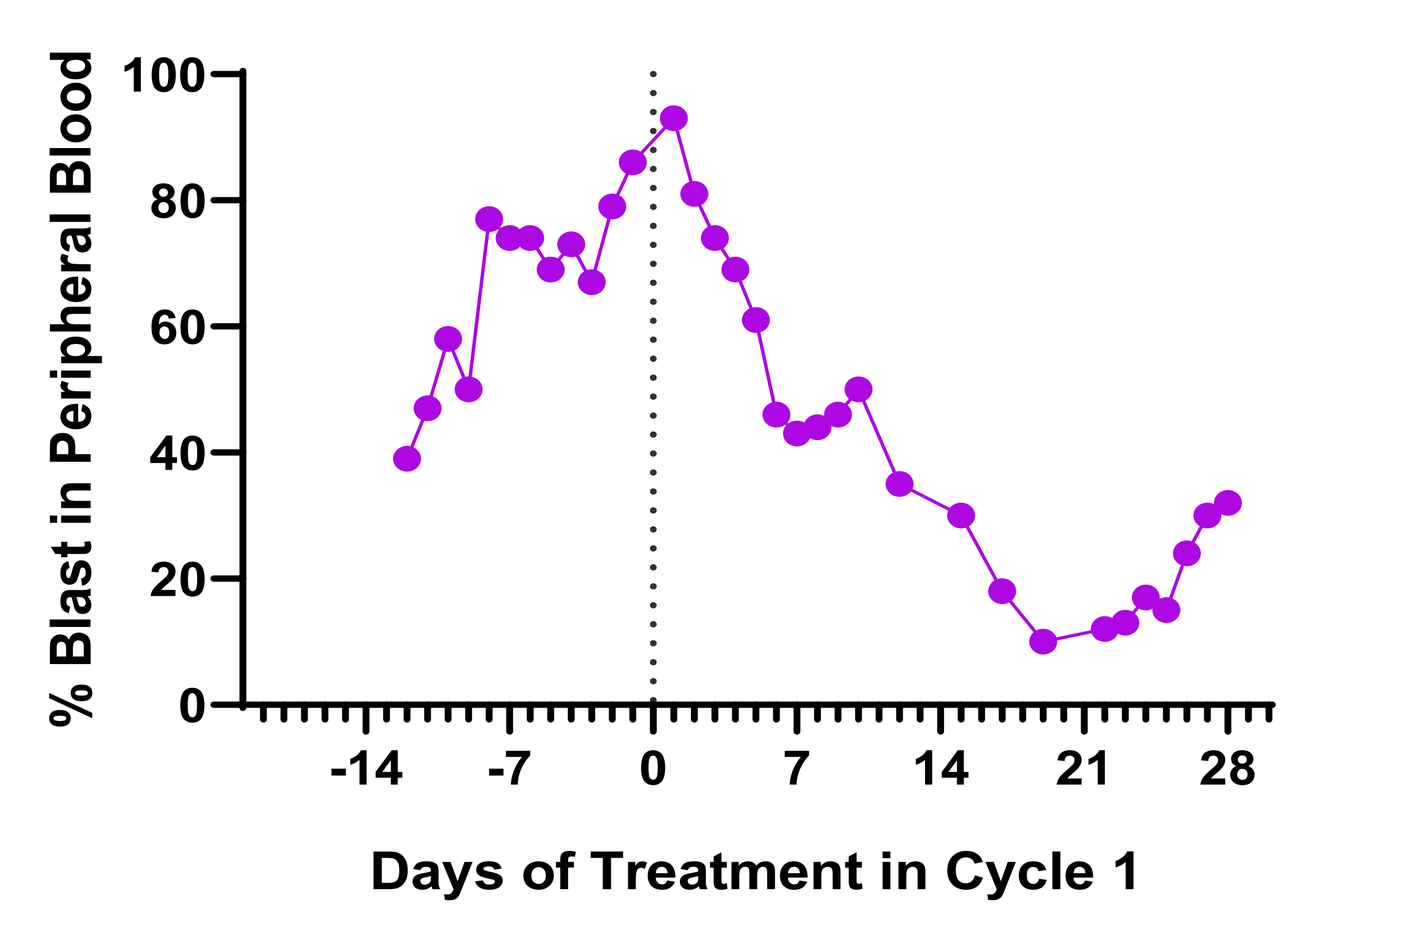
% Blast in Peripheral Blood
Days of Treatment in Cycle 1

Patient #3 - Heavily pretreated *de novo* AML: Lux effectively targeted FLT3-ITD clone

- 36y, female, treated with Lux 450mg BID
- **8 prior regimens** including FLT3 inhibitor gilteritinib and crenolanib, venetoclax and alloSCT
- Mutations detected at screening: FLT3-ITD, DNMT3A, NPM1, GATA2, WT1
- **Aggressively progressed before Lux treatment**
- **90+% reduction of blasts** in cycle 1, before disease progression in Cycle 2

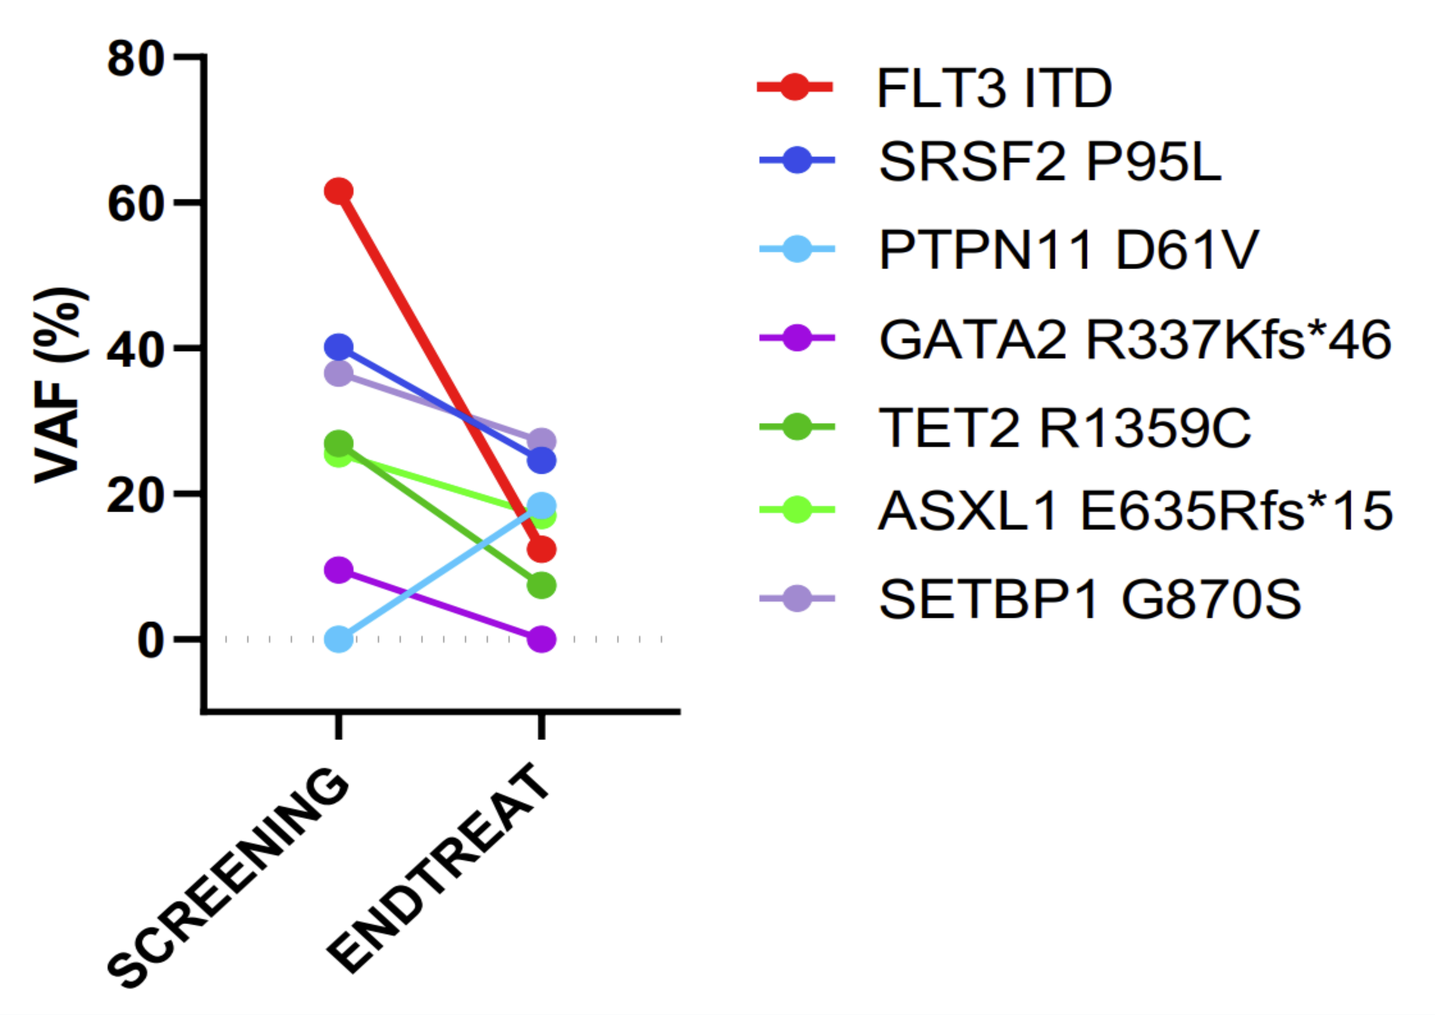
VAF (%)
FLT3 ITD
SRSF2 P95L
PTPN11 D61V
GATA2 R337Kfs*46
TET2 R1359C
ASXL1 E635Rfs*15
SETBP1 G870S
SCREENING
ENDTREAT

Patient #4 - *de novo* AML: Lux effectively targeted FLT3-ITD and other mutant clones associated with poor outcomes

- 76y, male, treated with Lux 450mg BID
- **2 prior regimens:** azacytidine, venetoclax
- **FLT3-ITD VAF (↓ 80%)** from 0.62 in peripheral blood at screening to 0.12 at the end of treatment (C4D9)
- Simultaneous reduction of ***GATA2* R337K (↓ 100%), *TET2* R1359C (↓ 73%), *SRSF2* P95L (↓ 39%) and *ASXL1* E635R (↓ 33%)**
- *PTPN11* mutant clone emerged at the end of treatment

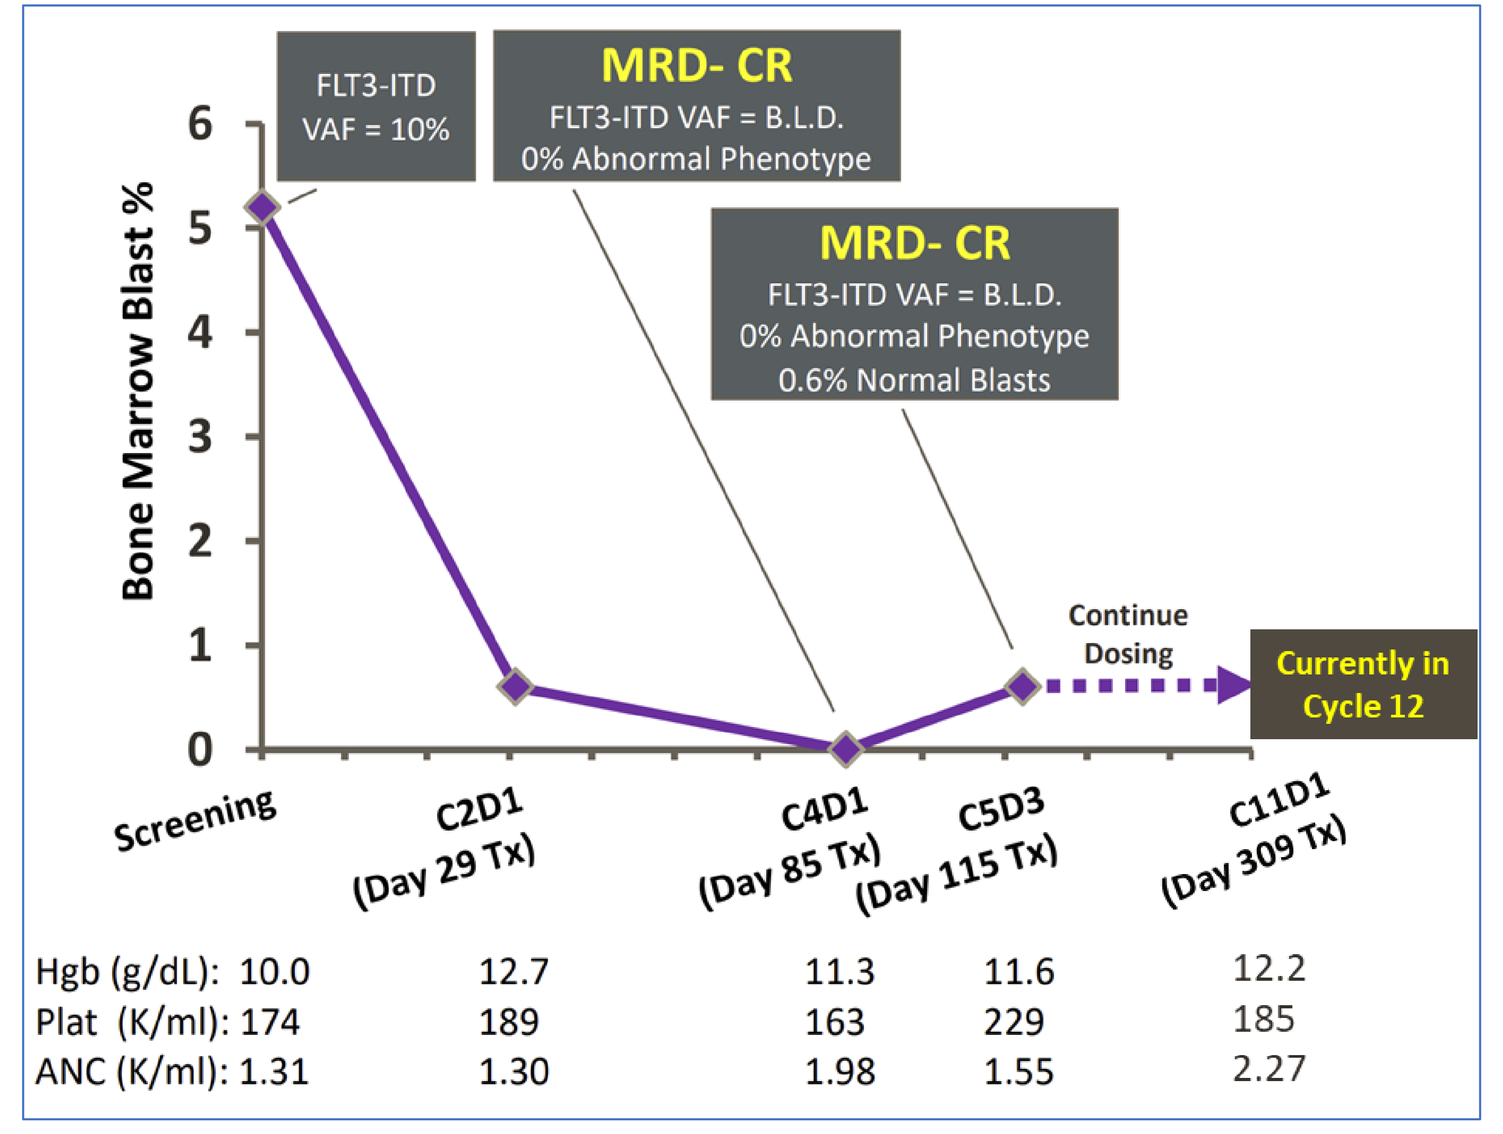
FLT3-ITD
VAF = 10%
MRD- CR
FLT3-ITD VAF = B.L.D.
0% Abnormal Phenotype
MRD- CR
FLT3-ITD VAF = B.L.D.
0% Abnormal Phenotype
0.6% Normal Blasts
Continue Dosing
Currently in Cycle 12
Bone Marrow Blast %
Screening
C2D1 (Day 29 Tx)
C4D1 (Day 85 Tx)
C5D3 (Day 115 Tx)
C11D1 (Day 309 Tx)
Hgb (g/dL): 10.0 12.7 11.3 11.6 12.2
Plat (K/ml): 174 189 163 229 185
ANC (K/ml): 1.31 1.30 1.98 1.55 2.27

Patient #5 – Heavily pretreated *de novo* AML / myeloid sarcoma: Lux eradicates FLT3-ITD clone and delivers MRD-negative CR

- 46y, male, treated with Lux 450mg BID
- **6 prior regimens** including FLT3 inhibitor sorafenib, venetoclax and 2 alloSCT
- **MRD-negative complete response** confirmed on C5D3 with FLT3-ITD VAF below detection limit (BLD) and blast< 0.1% by high-sensitivity flow cytometry
- **Patient continues on study in Cycle 12**

LUXEPTINIB PHASE 1a/b CONCLUSIONS

- **Luxeptinib is well tolerated at dose levels of 450, 600 and 750 mg BID over multiple cycles**
- **Two apparent DLTs (1 each in Cohorts 1 and 3) led to expansion; no DLTs occurred in the other 5 patients and supported dose escalation to next dose levels.**
- **PK plasma concentrations at steady state achieved ~1μM; PD biomarker documented inhibition of FLT3 signaling and other survival pathways**
- **Anti-leukemic activity has been observed in heavily pretreated relapsed FLT3-ITD AML patients as evidenced by significant reduction of FLT3-ITD VAF and blasts in bone marrow and / or peripheral blood.**
- **One FLT3-ITD AML patient has had confirmed MRD-negative CR and continues treatment in Cycle 12.**
- **Currently treating patients with R/R AML and higher-risk MDS at 900 mg BID in Cohort 4**
- **Currently also treating patients with R/R B-cell malignancies in a Ph 1 a/b study (NCT03893682, the latest update presented at ASH2021 abstract/poster#1355)**

Disclosures: Current clinical study is sponsored by Aptose Biosciences Inc. The following authors are employees of Aptose Biosciences Inc.: H Zhang, N Rastgoo, K Benbatoul, G Su, D Haney, Y Jin, J Marango, W Rice and R Bejar